DESIGN FOR A POPULATION STUDY OF EXPOSURE OF U.S. ADULT SMOKERS TO CIGARETTE SMOKE

R.-A. Walk[1], W.H. Carter[2], R.A. Carchman[2,3], J.C. Gear[1], W. Gomm[4], C. Gennings[2], R.D. Kinser[1], D.E. Leyden[5], B.L. Nelson[1], A.R.Tricker[6], K.von Holt[4]

[1]Philip Morris U.S.A., Richmond, VA, USA; [2]Department of Biostatistics, Virginia Commonwealth University, Richmond, VA, USA; [3]Columbia, VA, USA; [4]INBIFO Institit fuer biologische Forschung GmbH, Cologne, Germany; [5]Hartfield, VA, USA; [6]Philip Morris Products SA, Neuchatel, Switzerland*

WHY STUDY CIGARETTE SMOKE EXPOSURE?

KNOWN: Cigarette smoking causes lung cancer, heart disease, emphysema and other serious diseases in smokers

NEEDED: To determine the actual amount of internal (systemic) exposure of smoke constituents

WHY: To aid in the development and assessment of future reduced harm products

WHAT HAS BEEN DONE?

- **Machine Smoking Data** - Provide chemical smoke constituent measures in a standardized, reproducible fashion (i.e., U.S. Federal Trade Commission (FTC); International Organization for Standardization, Geneva (ISO); Massachusetts (U.S.) and Canada regulations)

 Concern: Does not provide exposure data that captures large range of smoking behaviors
- **Human Exposure Studies -** Indicate human smoke constituent exposure differs from that predicted by machine smoking data (e.g., Rosa et al., 1992; Byrd et al., 1995; Byrd et al., 1998; Djordjevic et al., 2000)

 Concerns: Typically single biomarker; based on small, non-representative samples; results not consistent among studies

WHAT NEEDS TO BE DONE?

Directly evaluate smoke constituents or their metabolites in appropriate biofluids ("biomarkers of exposure" and selected "biomarkers of effect")

- Identify relevant biomarkers of exposure (see Poster)
- Identify relevant biomarkers of effect
- Validate biomarkers for use in population studies
- Select representative population sample
- Examine the relationship between biomarkers of exposure and biomarkers of effect

WHAT WE PROPOSE

Conduct a *Total Exposure Study* (TES) representative of U.S. adult smokers of all tar yield (FTC) categories to generate baseline data for future exposure/harm reduction assessments and to determine whether smoke exposure differs for segments of FTC tar delivery (and non-smokers):

- in collaboration with, and carried out, by an independent contract research organization (CRO)
- large-scale
- multi-center
- cross-sectional

A *pilot study* is being conducted to determine the feasibility of the large-scale *Total Exposure Study* and determine estimates of variability for sample size determination.

EXCLUSION CRITERIA

- Persons less than 21 years of age (Photo id age verification required)
- Pregnant or nursing women
- Use of nicotine-containing tobacco & non-tobacco products other than manufactured cigarettes during past 3 months
- Diseases (diagnosis or manifestation of major chronic disease)
- Failure to pass *'healthy status tests'* (hematology, pulmonary function tests, vital signs and laboratory tests, renal insufficiencies, fever, and ECG)
- Participants on other clinical studies or blood donors/receivers
- Tobacco industry or CRO employees and immediate family

EXCLUSION CRITERIA SMOKERS

- Consumption of less than 1 manufactured cigarette of a specified brand per day for a minimum of 12 months
- Switched brands during last 3 months prior to enrollment
- Smoked a brand with tar delivery per cigarette different than the specified range within the last 3 months before enrollment
- Use of a different brand of cigarettes other than their preferred brand at a rate of more than 10% of daily consumption during the last three months prior to enrollment

EXCLUSION CRITERIA NON-SMOKERS

- History of smoking within the past 12 months
- History or use of any tobacco- or nicotine-containing products within 3 months prior to enrollment

COMPLEXITIES OF SAMPLING U.S. SMOKERS

For the Total Exposure Study (TES) to be representative of U.S. adult smokers of all tar yield (FTC) categories, the sample must reflect important demographic characteristics, such as:

- gender
- age
- race/ethnicity
- education
- income

Selection is further complicated when different tar levels are considered.

PILOT STUDY DESIGN

- Single-center
- Independent contract research organization (CRO)
- Healthy adult smokers of lower yield FTC "tar" cigarettes (3.0-6.9 mg tar)
- Non-smoking control group
- Feasibility study; 60 adult smokers, 60 adult non-smokers; 50% male/50% female

Objectives

- To establish the **validity of the design concepts** for the TES
- To determine the **intra- and inter-individual variability** of biomarkers of exposure and biomarkers of effect
- **To compare the biomarkers of exposure and the biomarkers of effects** for smokers and non-smokers

PILOT STUDY PARTICIPANTS

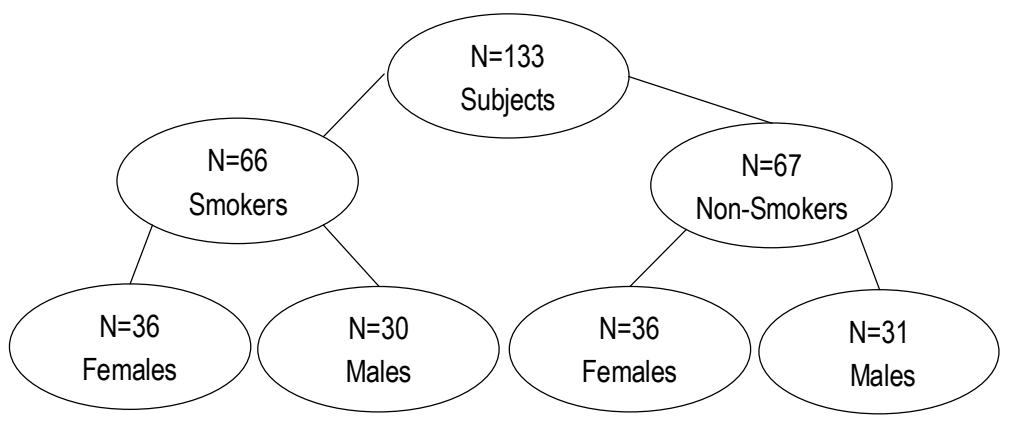
N=133
Subjects
N=66
Smokers
N=67
Non-Smokers
N=36
Females
N=30
Males
N=36
Females
N=31
Males

MEASUREMENTS

- Questionnaire, diary, weekly survey
 - Characterize population, smoking behavior and other exposures including environmental tobacco smoke
- Biomarkers of exposure
 - Quantify smoke exposure
- Biomarkers of effect
 - Assess potential harm

QUESTIONNAIRE

Characterize exposure to tobacco smoke and other exposures that could influence biomarker results:

- Verify subject inclusion
- Provide demographics (age, gender, ethnicity, marital status); smoking behavior characteristics (brand, number of cigarettes smoked/day, self-assessment of smoking behavior, etc.)
- Evaluate smoking behavior and allow for exposure characterization
- Provide occupational and other exposures (hobbies, home heating, engine exhaust)
- Examine self-reported exposure to cigarette smoke of others (strength and duration)
- Determine alcohol use; and physical activity
- Allow modeling of exposure and examination of outliers

BIOSAMPLES: PILOT

SAMPLE MATERIALS	BIOMARKER	WEEK OF SAMPLE COLLECTION*			
		1	2	3	6
Exhalate	Acetonitrile				
	Carbon monoxide				
Blood	Acetonitrile				
	COHb				
	3- and 4- ABP Hb adducts				
	LDL-, HDL-cholesterol				
	Malondialdehyde				
	Fibrinogen				
	C-reactive protein				
Urine	Nicotine and 5 metabolites				
	NNK metabolites (NNAL and NNAL-glucuronide)				
	11-Dehydro-thromboxane B_2				
	8-*epi*-prostaglandin $F_{2\alpha}$				
	Malondialdehyde				

*Sample collection made during the week indicated in gray

SUMMARY

- Design of *Total Exposure Study* is ongoing
- Pilot study is underway
- Data or suggestions are invited as input into this design

REFERENCES

Rosa, M. et al., How the steady-state cotinine concentration in cigarette smokers is directly related to nicotine intake. *Clin. Pharmacol. Ther.*, 1992:52; 324-9

Byrd, G.D., et al. Comparison of FTC-predicted nicotine uptake in smokers. *Psychopharmacology*, 1995:22; 95-103

Byrd, G.D., et al. A further study of FTC yield and nicotine absorption in smokers. *Psychopharmacology*, 1998:39; 291-299

Djordjevic, M. et al., Doses of nicotine and lung carcinogens delivered to cigarette smokers. *JNCI*, 2000:92(2): 106-11

Health Canada. Tobacco Legislation and Regulation. Tobacco Reporting Regulations. Available at http://www.hcsc.gc.ca/hppb/tobacco/ehd/tobacco/legislat/prop_may_36.htm. Accessed on June 20, 2001

105 CMR 660: Cigarette and smokeless tobacco products: Report of added constituents and nicotine ratings. Available at http://www.state.ma.us/dph/mtcp/report/nicdisc.htm. Accessed on June 20, 2001

*INBIFO is a Philip Morris research laboratory.