Lung Inflammation in Rats after Acute Exposure to Cigarette Smoke

P. Vanscheeuwijck[1], E. Van Miert[1], B. Friedrichs[2]; [1]PHILIP MORRIS Research Laboratories bvba, Leuven, Belgium; [2]PHILIP MORRIS Research Laboratories GmbH, Cologne, Germany

Patrick.Vanscheeuwijck@pmintl.com

Introduction

Last year (SOT, 2004) we reported that acute (1 day) inhalation exposure of rats to diluted cigarette mainstream smoke induces changes in inflammatory mediators in bronchoalveolar lavage fluid (BALF) and serum. Others have shown that upon exposing rodents to cigarette mainstream whole smoke (Ofulue et al., 1998; Obot et al., 2004), the number of neutrophils is increased in BALF. What is not known, however, is the relative contribution of the particulate phase and the gas phase of the smoke to pulmonary inflammation. Here, we investigate the inflammatory potential of cigarette mainstream whole smoke (WS), gas-phase-depleted particulate phase (PP), and gas phase (GP) in an acute and subchronic rat inhalation model.

Objective

- Investigate the relative contribution of PP and GP to pulmonary inflammation in an acute and subchronic rat smoke inhalation model.
- Determine whether acute inhalation exposure may be used as a short-term screening assay for the evaluation of the inflammatory effects of cigarette smoke.

Materials and Methods

Smoke Generation

- Generation of mainstream smoke according to ISO protocol (35 ml/puff in 2 s, each cigarette puffed once every minute, butt length 35 mm, Vanscheeuwijck et al., 2002)
 - WS collected from the smoking pump
 - PP generated by passing WS through an activated charcoal filter to remove the majority of gas phase constituents
 - GP generated by passing WS through an electrostatic filter to remove particulate matter

Test Atmosphere Characterization

- Concentrations of total particulate matter (TPM), carbon monoxide (CO), nicotine, and selected aldehydes (formaldehyde, acetaldehyde, acrolein) determined at the breathing zone of the animals

Rats and Treatment

Acute Inhalation Study
- Sprague-Dawley rats, 200 to 220 g at start of inhalation, 8 rats/group
- Nose-only exposure, 2 x 1 h with a 30-min break between the 2 h, 1 day only
- Exposure to fresh air (sham) or to mainstream smoke from the Kentucky Reference Cigarette 2R4F* at TPM concentrations of 300, 600, 900, and 1200 µg/l

Subchronic Inhalation Study
- Sprague-Dawley rats, 200 to 220 g at start of inhalation, 10 rats/group
- Nose-only exposure, 2 x 1 h/d with a 30-min break between the 2 h for 35 consecutive days
- Exposure to fresh air (sham) or to mainstream smoke from the Kentucky Reference Cigarette 1R4F* at TPM concentrations of 500 (WS only) and 750 µg/l

Sample Collection and Determinations

- BALF
 - approximately 20 h post-exposure
 - cannulation of isolated lungs via trachea
 - lavage with 5 consecutive cycles of filling (15 cm water pressure) and emptying (-8 cm water pressure)
 - filling medium: phosphate-buffered saline (PBS, Mg^{2+} and C^{2+} free) for the first cycle; PBS +0.3% bovine serum albumin (BSA) for cycles 2 to 5
 - determination of inflammatory mediators: in first cycle of lavage after centrifugation (cell-free)
 - free lung cell differentiation: cycles 2 to 5 combined, adjusted to 20,000 cells/ml, fixation with 2% formalin
- Inflammatory mediators
 - IL-1ß, TNFα, CINC-1, CINC-3, MCP-1, fractalkine using custom SearchLight Rat Cytokine/Chemokine Array (Pierce Biotechnology, Inc., Boston, MA)
- Free lung cell differentiation
 - staining: anti-granulocyte mAB-FITC (clone HIS48), anti-CD68-FITC (clone ED1), nucleic acid counterstaining using propidium iodide
 - flow cytometry using FACSVantage (BD Biosciences)—40,000 events/sample were counted
- Respiratory physiology parameters (6 rats/group)
 - head-out plethysmography
 - evaluation of respiratory minute volume (RMV) for each rat relative to pre-exposure value; data analysis IOX-software, EMKA Technologies

Statistics

- Results expressed as means ± SE
- One-way ANOVA followed by Dunnett test
- Differences considered statistically significant at p <0.05; asterisks in graphs indicate statistical differences compared to sham group

*2R4F is the remake of the 1R4F

Results

Acute Inhalation Study

Test Atmosphere Characterization

Group	TPM (µg/l)	CO (ppm)	Nicotine (µg/l)	Formald. (µg/l)	Acetald. (µg/l)	Acrolein (µg/l)
Sham	<QL	<QL	<QL	ND	ND	ND
WS 300	315	381	25.9	0.47	22.5	2.41
GP (300)	<QL	340	0.24	0.20	21.6	1.76
PP 300	285	343	18.5	0.14	8.6	0.25
WS 600	632	645	47.8	0.80	46.2	4.72
GP (600)	<QL	652	0.11	0.38	44.7	4.23
PP 600	609	699	37.5	0.27	11.7	0.45
WS 900	896	830	65.7	1.12	59.5	6.15
GP (900)	<QL	981	0.19	0.53	71.4	6.72
PP 900	910	1051	52.3	0.43	12.6	0.50
WS 1200	1179	1158	81.3	1.23	77.0	7.57
GP (1200)	<QL	1355	<QL	0.49	97.3	8.27
PP 1200	1151	1371	62.7	0.53	25.1	0.83

QL = quantitation limit. For TPM: QL = 12 µg/l; for CO: QL = 5 ppm; for nicotine: 0.1 µg/l
ND = not determined; n = 1 for all parameters

CINC-3

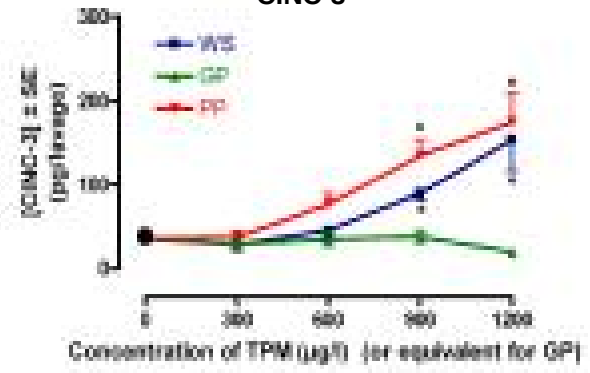
WS
GP
PP
[CINC-3] ± SE (pg/lavage)
Concentration of TPM (µg/l) (or equivalent for GP)

Fractalkine

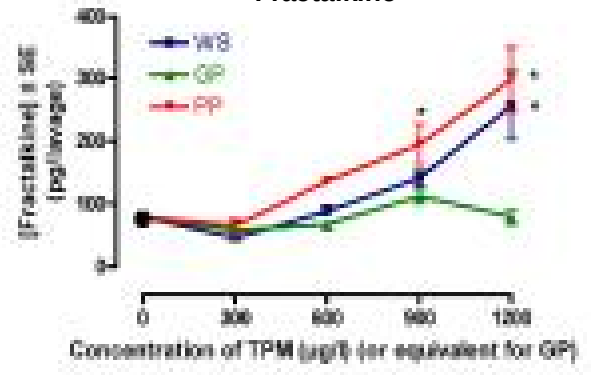
WS
GP
PP
[Fractalkine] ± SE (pg/lavage)
Concentration of TPM (µg/l) (or equivalent for GP)

Inflammatory Mediators in BALF
- Cytokines and chemokines increased after exposure to WS
- Most pronounced effect after exposure to PP
- No effect after exposure to GP
- Similar response seen for CINC-1 and MCP-1 (data not shown)
- No response seen for TNFα (data not shown)

IL-1b

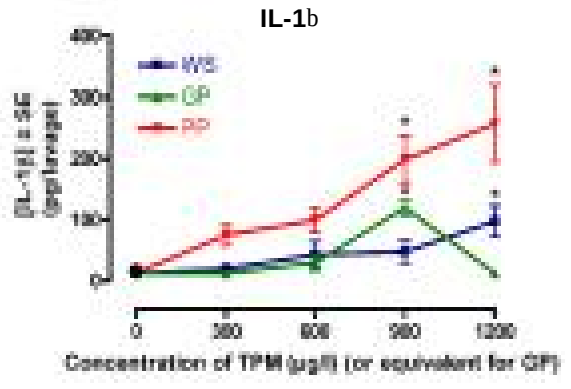
WS
GP
PP
[IL-1β] ± SE (pg/lavage)
Concentration of TPM (µg/l) (or equivalent for GP)

Neutrophils in BALF
- Neutrophils increased after exposure to WS
- Most pronounced effect after exposure to PP
- No effect after exposure to GP

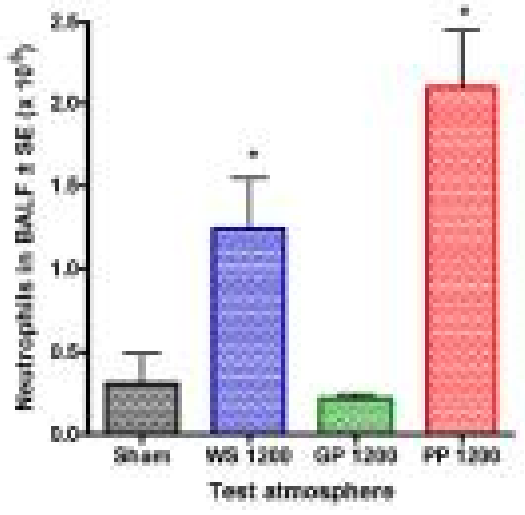
Neutrophils in BALF ± SE (x 10^5)
Sham
WS 1200
GP 1200
PP 1200
Test atmosphere

Subchronic Inhalation Study

Test Atmosphere Characterization

Group	TPM (µg/l)	CO (ppm)	Nicotine (µg/l)	Formald. (µg/l)	Acetald. (µg/l)	Acrolein (µg/l)
Sham	< QL	< QL	< QL	ND	ND	ND
WS 500	514 ± 25	538 ± 30	36.0 ± 3.1	0.6 ± 0.1	22.3 ± 2.0	1.5 ± 0.1
WS 750	759 ± 49	746 ± 43	50.6 ± 5.4	0.9 ± 0.2	32.0 ± 4.1	2.1 ± 0.3
GP (750)	< QL	788 ± 67	< QL	0.2 ± 0.1	31.0 ± 4.8	1.8 ± 0.4
PP 750	744 ± 61	913 ± 78	42.7 ± 4.7	0.3 ± 0.1	10.2 ± 7.0	0.2 ± 0.0

QL = quantitation limit. For TPM: QL = 12 µg/l; for CO: QL = 5 ppm; for nicotine: 0.1 µg/l
ND = not determined; n = 35 for TPM and CO, n = 5 for nicotine and aldehydes; means ± SD

Neutrophils in BALF
- Neutrophils increased after exposure to WS
- Most pronounced effect after exposure to PP
- No effect after exposure to GP

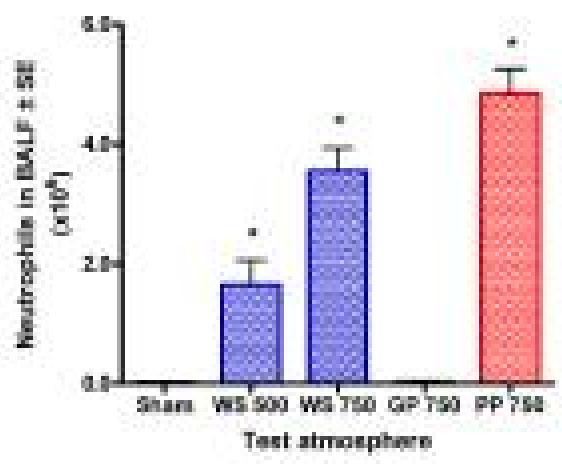
Neutrophils in BALF ± SE (x10^6)
Sham
WS 500
WS 750
GP 750
PP 750
Test atmosphere

Respiratory Minute Volume
- RMV depressed during exposure to WS and GP
- No depression during exposure to PP

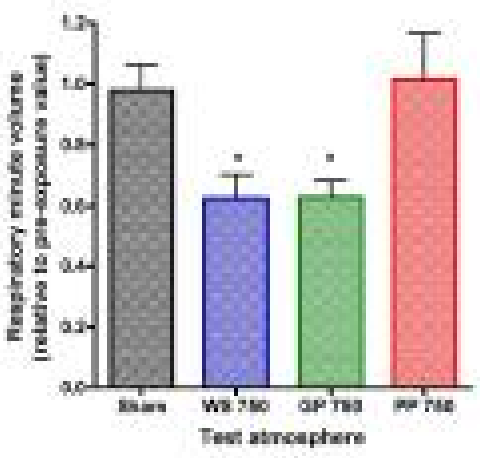
Sham
WS 750
GP 750
PP 750
Test atmosphere

Summary and Discussion

- WS induces inflammatory changes in BALF from rats after acute and subchronic inhalation exposure.
- PP is responsible for the inflammatory changes in the rat lung, GP had no effect.
- PP induces qualitatively the same inflammatory changes as WS, but to a greater extent. This is most likely due to the higher uptake as indicated by the lack of depression of respiratory minute volume in the rats exposed to PP.
- Inflammatory changes in the rat lung after acute inhalation exposure parallel the changes observed after subchronic inhalation exposure.

Conclusion

- Mainstream whole smoke and gas-phase-depleted particulate phase cause similar inflammatory changes in the rat lung.
- Acute inhalation exposure may be a useful short-term in vivo assay for the evaluation of the inflammatory effects of cigarette smoke.

References
Ofulue, A.F., Ko, M., Abboud, R.T., Am. J. Physiol. 275 (1998) L1134-L1144.
Obot, C.J., Lee, K.M., Fuciarelli, A.F., Renne, R.A., McKinney, W.J., Inhal. Toxicol. 16 (2004) 701-719.
Vanscheeuwijck, P., Van Miert, E., Kuhl., P, abstract no. 1277, 2004.
Vanscheeuwijck, P., Teredesai, A., Terpstra, P.M., Verbeeck, J., Kuhl, P., Gerstenberg, B., Gebel, S., Carmines, E.L., Food and Chem. Toxicol. 40 (5) (2002) 113-131.

3rd Siena International Conference on Animal Models of Chronic Obstructive Pulmonary Disease
Siena, 24 to 26 September 2005 (also presented at SOT 2005)