Detection of $CD4^+$ and $CD8^+$ T-Lymphocytes in Lung Tissue of A/J-Mice by Immunohistology

A. Büttner, S. Töller, T. Grün, H. Weiler
PHILIP MORRIS Research Laboratories GmbH, 51149 Cologne, Germany

Ansgar.Buettner@pmintl.com

Introduction

$CD4^+$ T-lymphocytes (T-helper/inducer cells) and $CD8^+$ T-lymphocytes (T-suppressor/cytotoxic cells) are regarded as important cells in the pathogenesis of cigarette smoke-induced lung diseases (COPD, emphysema, asthma) in humans (Saetta et al., 1998; Takubo et al., 2002; Guerassimov et al., 2004). To allow investigation of a possible shift in the relative numbers of $CD4^+$ and $CD8^+$ T-lymphocytes after cigarette smoke inhalation, as seen in the human situation, we adapted immunohistological procedures as documented in the literature to our needs and established a method for staining the different T-lymphocyte subpopulations in lungs of A/J-mice thus enabling quantification of $CD4^+$/$CD8^+$ T-lymphocytes.

Methods

Animals/Tissue

The lung and the spleen of untreated male A/J-mice, bred under specific pathogen-free conditions (The Jackson Laboratory, Bar Harbor, Maine, U.S.A.) were taken for the establishment of the immunohistological staining protocol. The spleen was used as positive control where T-lymphocytes are located in the periateriolar lymphoid sheath (PALS) of the white pulp.

Fixation and Slide Processing

The tissue for immunohistological investigation was fixed immediately after dissection with HOPE (Hepes Glutamic Acid Buffer Mediated Organic Solvent Protection Effect) (DCS Innovative Diagnostic System, Hamburg, Germany) solution according to a standard protocol. The lung was instillated via the trachea with the fixative under constant pressure of 20 cm H_2O. After paraffin-embedding, histological slides of 4 µm thickness were prepared.

Staining Procedure

Rat monoclonal antibodies were used for identification of $CD4^+$ (Rt anti-Ms, Clone YTS191.1, Serotec) and $CD8^+$ (Rt anti-Ms, Clone KT15, Serotec) lymphocytes. Monoclonal antibody binding was detected with the ABC-kit for peroxidase reaction (PK-6102, Vektor) and Diaminobenzidin (Dako) substrate.

Results

Spleen: An intensive cell specific brown to yellow cytoplasmatic staining of lymphoid cells mainly in the PALS of the white pulp can be seen after incubation with CD4 and CD8 antibodies (**Pictures 1** and **2**). Few scattered lymphoid cells in the red pulp and in the follicular center react positive.

Lung: In the lung tissue positive stained cells were mainly located in the alveolar walls. The staining result after incubation with CD8 was slightly more intensive than after incubation with the CD4 antibody (**Pictures 3** and **4**).

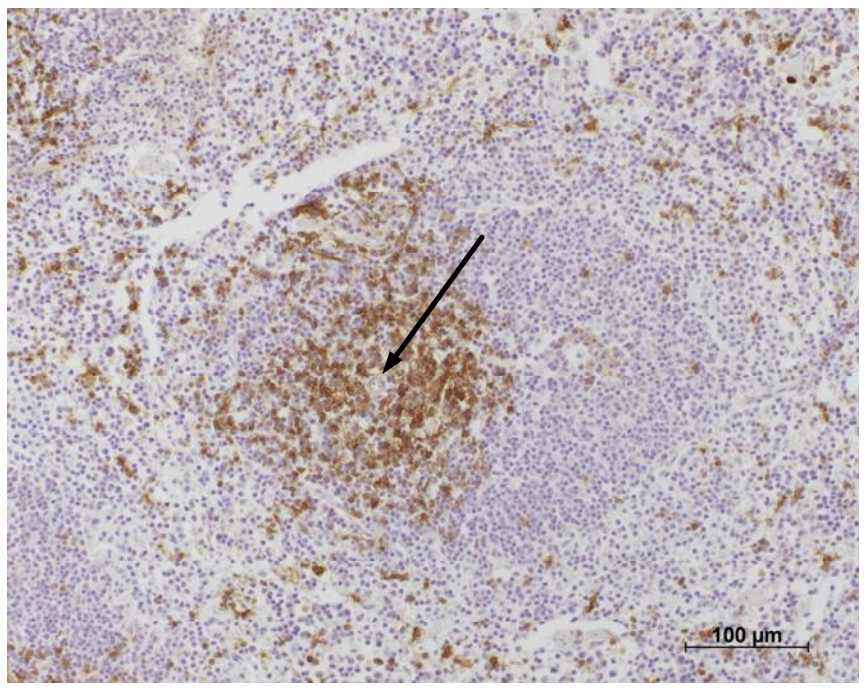
100 µm

Picture 1: Spleen, mice, anti CD8-antibody, stained lymphoid cells are mainly located at the PALS (arrow), ABC-complex, peroxidase reaction, Diaminobenzidin, 200x

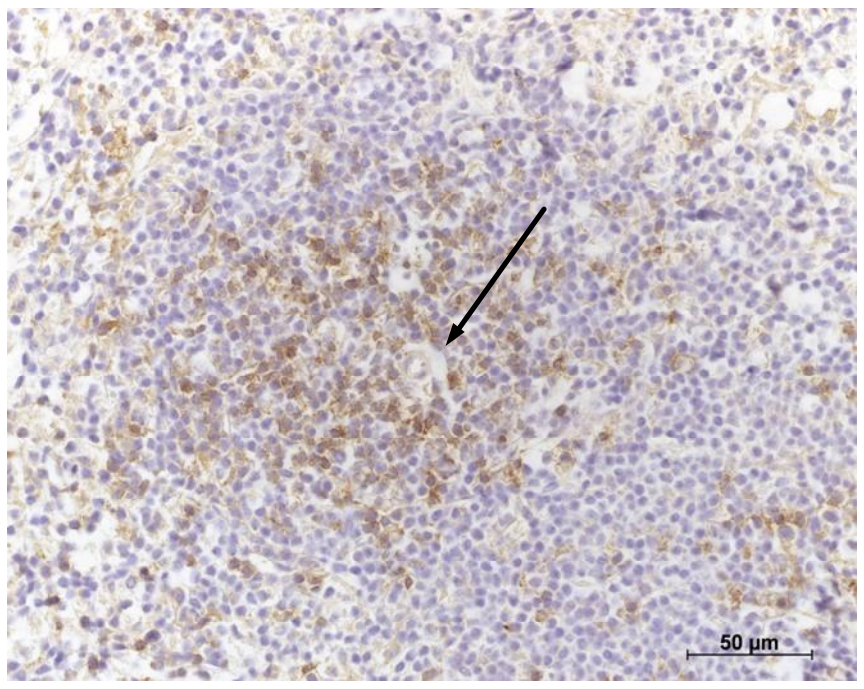
50 µm

Picture 2: Spleen, mice, anti CD4-antibody , stained lymphoid cells are mainly located at the PALS (arrow), ABC-complex, peroxidase reaction, Diaminobenzidin, 400x

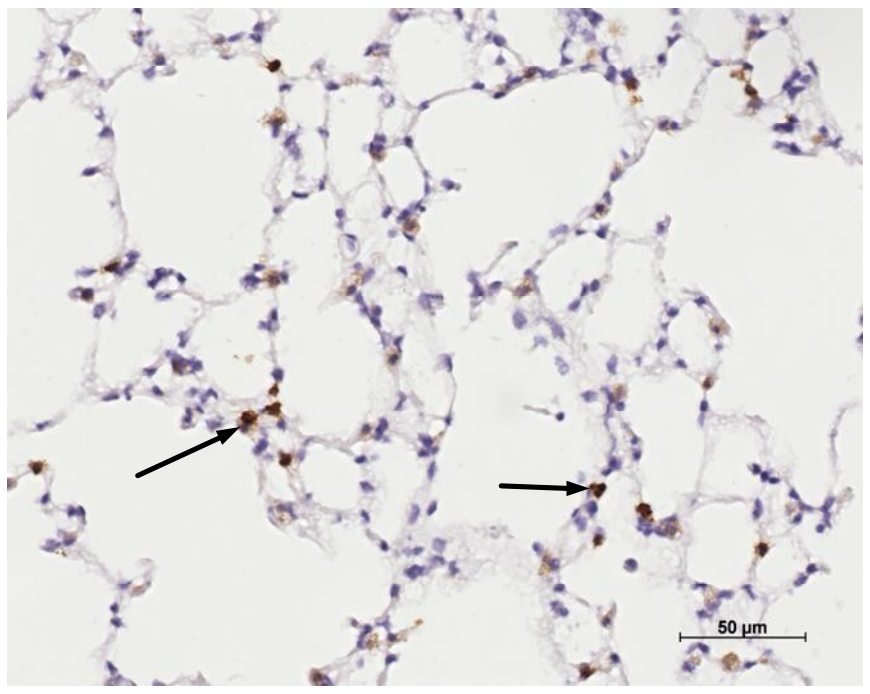
50 µm

Picture 3: Lung, mice, anti CD8-antibody, stained cells are located inside the alveolar wall (arrow), ABC-complex, peroxidase reaction, Diaminobenzidin, 400x

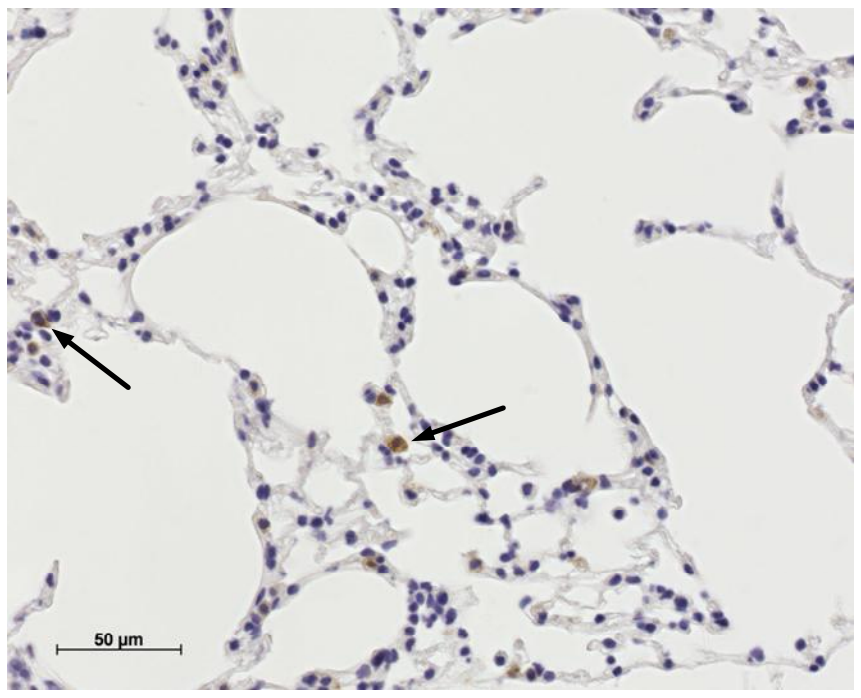
50 µm

Picture 4: Lung, mice, anti CD4-antibody, stained cells are located inside the alveolar wall (arrow). The amount of positive cells is slightly lower compared with the results after incubation with CD8 antibody. ABC-complex, peroxidase reaction, Diaminobenzidin, 400x

Discussion

For establishment of an immunohistological detection system of T-cell subpopulations, monoclonal antibodies against the CD4 and CD8 antigens were used. Using spleen tissue as the positive control, a specific staining of T-lymphocytes with both antibodies can be shown. The staining results from the lung tissue might allow the quantification of each cell population. The technique of immunohistological characterization of $CD4^+$ and $CD8^+$ lymphocytes in lung tissue might be a useful method to investigate the influence of T-lymphocytes during the development of lung diseases after inhalation of cigarette smoke in A/J-mice.

X:\ABU_2005_HSR.vsd

References

Guerrassimov, A., Hoshino, Y., Takubo, Y., Turcotte, A., Yamamoto, M., Ghezzo, H., Triantafillopoulos, A., Whittaker, K., Hoidal, J.R., Cosio, M.G., The development of emphysema in cigarette smoke-exposed mice is strain dependent, Am. J. Respir. Crit. Care Med. 170 (9): 974-980 (2004)

Di Stefano, A., Caramori, G., Ricciardolo, F.L., Capelli, A., Adcock, I.M., Donner, C.F., Cellular and molecular mechanisms in chronic obstructive pulmonary disease: an overview, Clin. Exp. Allergy 34 (8): 1156-1167 (2004)

Takubo, Y., Guerrasimov, A., Ghezzo, H., Triantafillopoulos, A., Bates, J.H.T., Hoidal, J.R., Cosio, M.G., α_1-Antitrypsin determines the pattern of emphysema and function in tobacco smoke-exposed mice: parallels with human disease, Am. J. Respir. Crit. Care Med. 166 (12 I): 1596-1603 (2002)

Saetta, M., Di Stefano, A., Turato, G., Facchini, F.M., Corbino, L., Mapp, C.E., Maestrelli, P., Ciaccia, A., Fabbri, L.M., $CD8^+$ T-Lymphocytes in peripheral airways of smokers with chronic obstructive pulmonary disease, Am. J. Respir. Crit. Care Med. 157 (3): 822-826 (1998)

presented at the Health Sciences Research Information Sharing Meeting, 2-4 November 2005, Lansdowne Resort, Lansdowne, VA, U.S.A.