Analysis of the extent of emphysema in live mice by high resolution X-ray microtomography

Andrei A. Postnov[1,2], Kris Meurrens[3], Horst Weiler[4], Dirk Van Dyck[2] , Haiyan Xu[3], Piter Terpstra[3], and Nora M. De Clerck[1]

[1]Department of Biomedical Sciences, [2]Department of Physics, University of Antwerp, Universiteitsplein 1, B-2610 Antwerp, Belgium

[3,4]PHILIP MORRIS Research Laboratories, [3]Grauwmeer 14, B-3001 Leuven, Belgium and [4] Fuggerstrasse 3, D-51149 Köln, Germany

Emphysema has been defined as the permanent enlargement of air spaces distal to the terminal bronchiole, caused by destruction of alveolar walls and without significant fibrosis. As emphysema is based on an anatomical derangement, imaging techniques can be considered as a useful tool for the detection of this disease in animals. For an *in vivo* assessment of emphysema non-invasive imaging techniques are required. Therefore, the major purpose of the present pilot study was to analyze whether high resolution X-ray microtomography is able to detect the degree of emphysema in live mice. Lungs of healthy mice were compared with lungs affected by different degrees of emphysema.

Sixteen male, 8-week-old, C57BL/6J mice (Charles River, Germany), mean bodyweight 22.3 g *[+/- 0.3 (SE)]*, were assigned to four treatment groups. The four mice in each group were given intratracheally 50µl physiological saline alone (= control vehicle) or a solution of 0,75 (low dose), 1,5 (medium dose) and 3,0 (high dose) mg/kg porcine pancreatic elastase (PPE, 281 U/mg, Serva, Germany) in saline.

For scanning, an *in vivo* X-ray micro-CT system was used (Skyscan 1076, Aartselaar, Belgium) without gating for cardiac or respiratory motion. Both the X-ray source (focal spot size 5 µm, energy range 20-100keV) and the detector (CCD camera 2.3kx4k) rotated around the animal. www.skyscan.be.

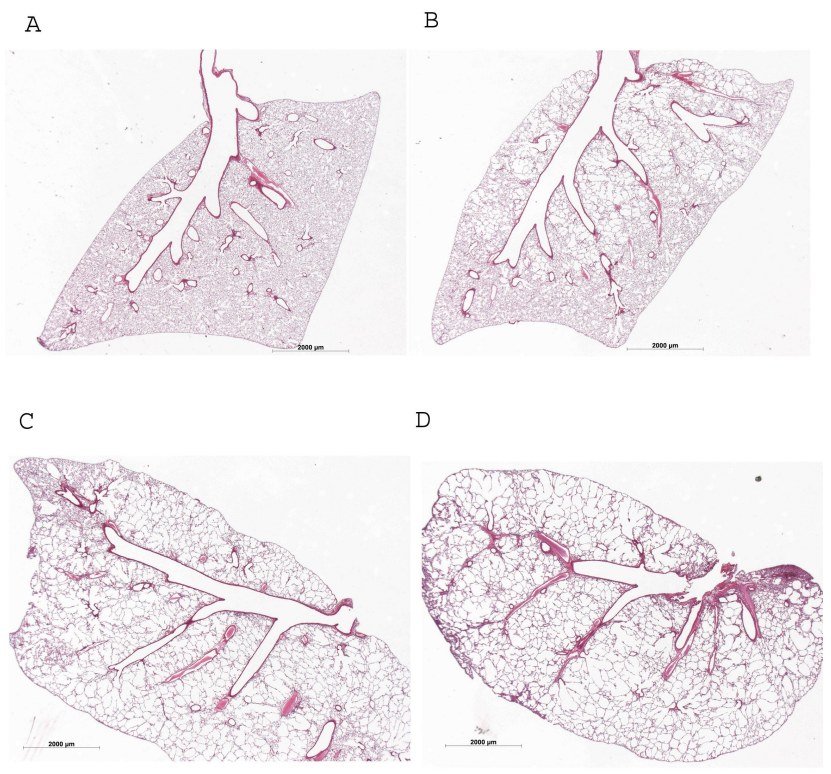
A
B
C
D

Figure 1: Histopathological evaluation of the lungs of the elastase-treated mice.
Different levels of emphysema are indicated.
Panel A: control, panel B: low dose, panel C: medium dose, panel D: high dose of elastase treatment

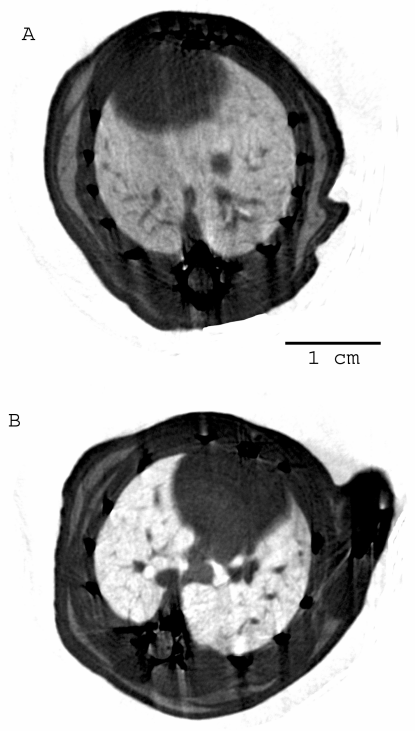
A
1 cm
B

Figure 2: Representative virtual cross-section through the chest area of a control mouse (panel A) and of a mouse from the high dose group (panel B). Reconstruction parameters were the same in both cross-sections. Differences in density of the lungs are obvious.

Figure 3: The frequency of spatial occurrence of each CT number versus the absolute HU values. The lungs were selected as the region of interest. Panel A: total frequency distribution of the CT numbers in all cross-sections (range -200-830 HU) versus the CT numbers. Panel B: individual dots represent tissue lighter than –830 HU. Appearance of CT numbers corresponded in a dose dependent way to the elastase treatment.

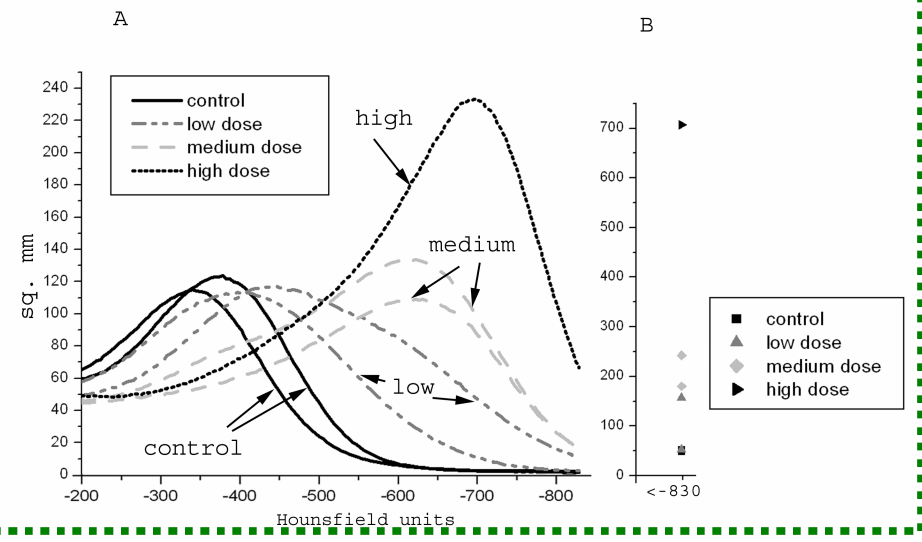
A
B
control
low dose
medium dose
high dose
high
medium
low
control
sq. mm
Hounsfield units
<-830

E-mail: andrei.postnov@ua.ac.be

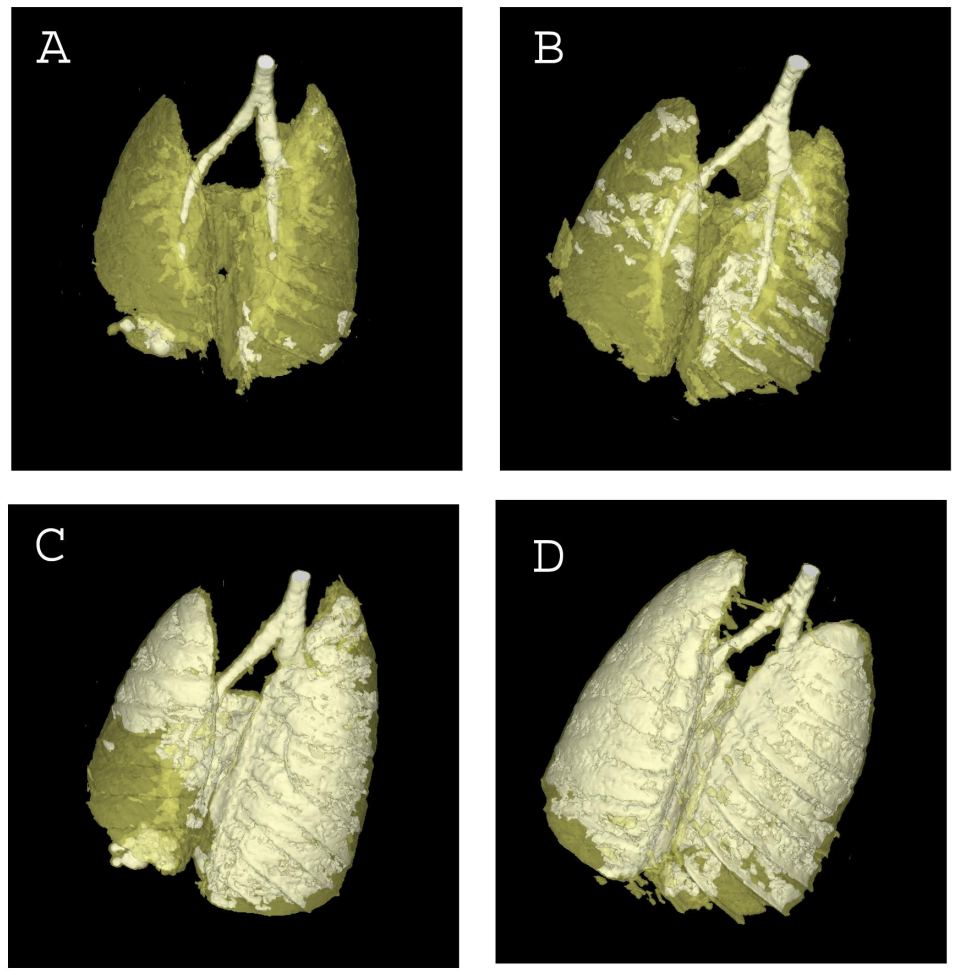
A
B
C
D

Figure 4: Three-dimensional models of the entire lungs, with superposed on them three-dimensional models of the air (void space). Panel A : control; Panel B : low dose, Panel C: medium dose and Panel D : high dose. The high dose group has the largest empty space, and the largest volume.

Group	Lung volume (mm³)	Tissue with CT numbers < -620HU (%)*
Control	507, 506	1.9, 2.4
Low dose	607, 810***	4.4, 15.9***
Medium dose	839, 768	29.2, 28.9
High dose**	1218	51.5

Table 1 : Quantification of the degree of emphysema related to the entire lung volume: column 2 represents total lung volume, in column 3 the volume occupied by void space and affected tissue is indicated (i.e. tissues with CT numbers less than –620HU).

(*) The threshold for the visualization of emphysema was chosen to clearly distinguish affected from normal tissue. In control animals some percentage of void space is due to bronchial airways containing air (no X-ray absorption).

(**) Only one animal survived in high dose group.

(***) One mouse from the low dose group turned out to be more affected than expected.