Inflammatory and Epithelial Changes in Lungs from Apolipoprotein-E-Deficient Mice after Chronic Cigarette Mainstream Smoke Exposure

S. Lebrun[1], W. Stinn[1], H. Weiler[1], P. Kuhl[1], B. Friedrichs[1], A. Büttner[1], K. von Holt[1], T. Wallerath[1], R. Schleef [2], and H.-J. Haussmann[3]
[1] PHILIP MORRIS Research Laboratories GmbH, Fuggerstraße 3, 51149 Cologne, Germany; [2] PHILIP MORRIS USA, Richmond, VA, USA; [3] Rösrath, Germany
Stefan.Lebrun@pmintl.com

Introduction

Cigarette smoke (CS) exposure induces oxidative stress and inflammatory response in mice and humans alike. The biochemical alterations induce functional and morphologic changes thereby ultimately increasing the risk for the development of cancer, chronic obstructive pulmonary disease, and atherosclerosis[1].

Specifically, several studies in mice have shown that CS exposure leads to increased numbers of macrophages, neutrophils, and lymphocytes in bronchoalveolar lavage fluid (BALF) in mice[2-4]. D'hulst et al. reported a mixture of acute and chronic inflammatory changes in mice exposed to CS for 6 months. These changes are the result of a combination of innate and adaptive immune responses to CS; all augmented cell types are involved in both types of immune response[4].

As part of a study on the influence of cigarette mainstream smoke (MS) in combination with different diets on the development of atherosclerosis[5], we investigated the effects of MS on lungs of apolipoprotein-E-deficient mice (ApoE$^{-/-}$). This mouse model develops atherosclerosis, a chronic inflammatory disease of large and medium arteries[6].

Objective

Investigate the inflammatory response and histopathological alterations in lungs from ApoE$^{-/-}$ mice exposed to MS and fed a normal chow or milk-fat-enriched diet.

Materials and Methods

Smoke Generation
- 2R4F (University of Kentucky Reference Cigarette), conditioning according to ISO standard 3402[7]
- MS generation on a modified automatic 30-port smoking machine (type: INBIFO SM85) equipped with a 4-piston Battelle pump[8] according to ISO standard 3308[9]

Animals and Treatment
- Male ApoE$^{-/-}$ mice (C57BL/6-apo Etm1Unc, Charles River Laboratories, France)
- Care and use of the mice was in conformity with AALAS policy[10]
- 20 mice per group and time point
- Whole-body exposure, 6 hours/day, 5 days/week
- Exposure to filtered fresh air (sham) or to MS at total particulate matter (TPM) concentrations of 100 µg/l and 200 µg/l
- Normal chow diet (0.02% cholesterol/4.5% fat) or milk-fat-enriched diet (0.17% cholesterol/21% fat) ad libitum
- Dissection and sample preparation after 3, 6, 9, or 12 months of exposure

Carboxyhemoglobin (COHb)
- Collection of blood samples from retrobulbar venous plexus of halothane-anesthetized mice during the last hour of exposure
- COHb determination using gas chromatography[8]

Bronchoalveolar Lavage (BAL) Cell Differentiation (8 mice/group)
- BAL hours after last exposure, 7 cycles with Mg^{2+}/Ca^{2+}-free phosphate-buffered saline + 0.325% bovine serum albumin, filling at 15 cm water pressure, emptying at −8 cm water pressure
- Fixation of cells in 2% formalin
- Staining with anti-neutrophil mAB-FITC (clone7/4)
- Nucleic acid counterstaining with propidium iodide
- Flow cytometry using FACSVantage, BD Biosciences

Histopathological Evaluation (12 mice/group)
- Instillation of lung tissue with 4% formaldehyde solution
- Embedding in paraplast, sectioning according to standard level (main bronchus)
- Hematoxylin/eosin staining (H&E)
- Semiquantitative evaluation (scoring system 0 to 5) of pigmented/non-pigmented macrophages by conventional light microscopy
- Incidence of pigmented macrophage nests (PMN), pneumocyte hypertrophy/hyperplasia and lymphocytic infiltrates was determined by conventional light microscopy

Statistics
- One-way ANOVA followed by Dunnett
- Data presented as means ± SE; *: p ≤0.05

Results

Body Weight Development

Body weight was up to 10% lower in the smoke-exposed groups compared to sham (normal chow diet only).

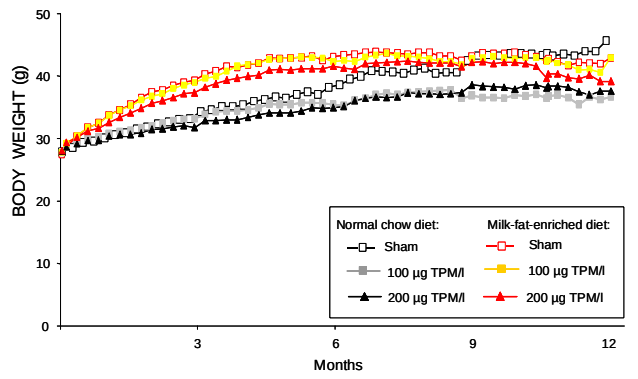
BODY WEIGHT (g)
Months
Normal chow diet:
Sham
100 µg TPM/l
200 µg TPM/l
Milk-fat-enriched diet:
Sham
100 µg TPM/l
200 µg TPM/l

Carboxyhemoglobin

Carboxyhemoglobin levels were 11% ± 0.3 in the groups exposed to 100 µg TPM/l MS and 20% ± 0.6 in the groups exposed to 200 µg TPM/l MS.

Pigmented Macrophage Nests (PMN)

H&E-stained lung slice from an ApoE$^{-/-}$ mouse fed a milk-fat-enriched diet and exposed for 12 months to 200 µg TPM/MS. Arrows indicate PMN.

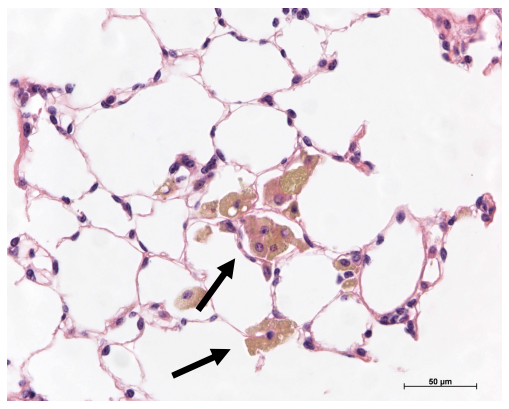
50 µm

Hypertrophy/Hyperplasia of the Alveolar Epithelium

H&E-stained lung slice from an ApoE$^{-/-}$ mouse fed a milk-fat-enriched diet and exposed for 12 months to 200 µg TPM/l MS. Arrows indicate hyperplasia of the alveolar epithelium associated with PMN.

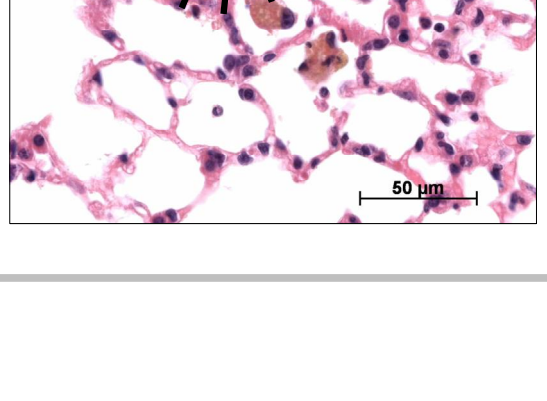
50 µm

Inflammatory Cells in BALF

ApoE$^{-/-}$ mice exposed to 200 µg TPM/l MS showed a constant elevation of neutrophils and a continuous increase in lymphocytes in BAL cells in both diet groups. In the milk-fat-enriched diet groups, there was a tendency to a greater increase in neutrophils and lymphocytes.

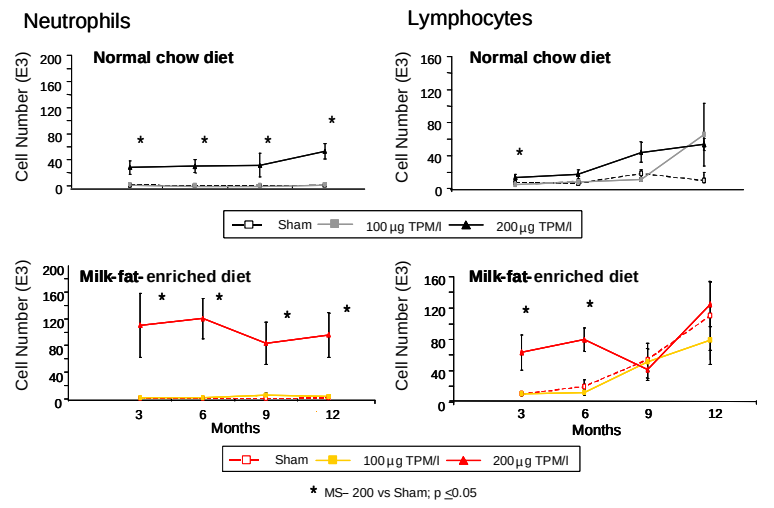
Neutrophils
Lymphocytes
Normal chow diet
Milk-fat-enriched diet
Cell Number (E3)
Months
Sham
100 µg TPM/l
200 µg TPM/l
* MS- 200 vs Sham; p ≤0.05

Pigmented and Non-Pigmented Alveolar Macrophages

Exposure to 200 µg TPM/l MS resulted in a continuous increase in pigmented and non-pigmented alveolar macrophages in lungs of ApoE$^{-/-}$ mice in both diet groups. Mean scores increased from 0.5 (3 months) to 1.7 (12 months).

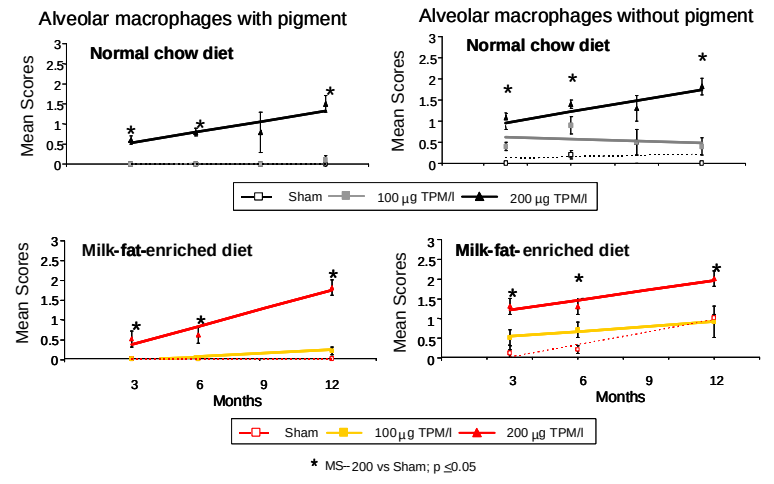
Alveolar macrophages with pigment
Alveolar macrophages without pigment
Normal chow diet
Milk-fat-enriched diet
Mean Scores
Months
Sham
100 µg TPM/l
200 µg TPM/l
* MS- 200 vs Sham; p ≤0.05

Incidence of Hypertrophy/Hyperplasia of Alveolar Epithelium and Incidence of PMN

The incidence of hypertrophy/hyperplasia and PMN in ApoE$^{-/-}$ mice exposed to 200 µg TPM/l MS increased from 20% (3 months) to 80% (12 months) in both diet groups. Hypertrophy/hyperplasia of the alveolar epithelium was seen only in single cells directly associated with the PMN.

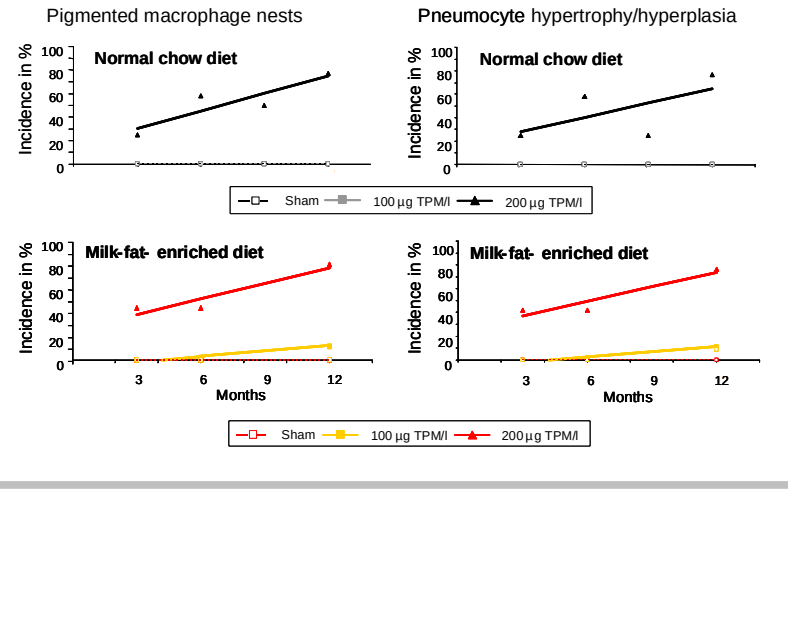
Pigmented macrophage nests
Pneumocyte hypertrophy/hyperplasia
Normal chow diet
Milk-fat- enriched diet
Incidence in %
Months
Sham
100 µg TPM/l
200 µg TPM/l

Interstitial Lymphocytic Infiltrates

(A) Representative image of H&E-stained lung slice from an ApoE$^{-/-}$ mouse fed a normal chow diet and exposed for 12 months to 200 µg TPM/l. Arrows indicate interstitial lymphocytic infiltrates.

(B) Interstitial lymphocytic infiltrates were present in 80% of ApoE$^{-/-}$ mice exposed to 200 µg TPM/l MS in both diet groups only after 12 months of exposure.

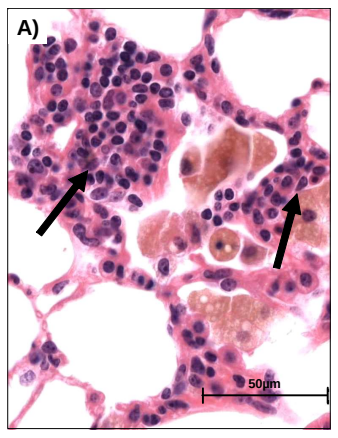
A)
50µm

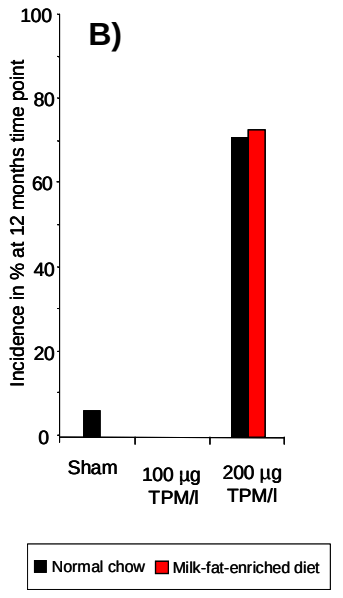
B)
Incidence in % at 12 months time point
Sham
100 µg TPM/l
200 µg TPM/l
Normal chow
Milk-fat-enriched diet

Conclusion

These data indicate that chronic exposure of ApoE$^{-/-}$ mice to 200 µg TPM/l MS (but not to 100 µg TPM/l MS) resulted in non-neoplastic pathomorphological changes in the lung and an inflammatory response. The inflammatory response is a combination of innate and adaptive immune responses, as indicated by an increase in BAL neutrophils and lymphocytes.

Diet type had little effect. The inflammatory response was only marginally modulated by the milk-fat-enriched diet; histopathological alterations in the lung were not influenced by diet.

Acknowledgements

The authors thank Christina Bajraktari, Andrea Daniels, Stefanie Gaschina, Thomas Grün, Renate Janssens, Oliver Jörke, Sabine Jurgeit, Kerstin Landwehr, Maik Lorbeer, Nina Plewka, Daniela Sampels, and Andrea Völkel for their excellent technical assistance.

References

1. U.S. Department of Health and Human Services, Rockville, MD, 2004
2. Seagrave, J., Barr, E.B., March, T.H., Nikula, K.J., Exp. Lung Res. 30(1): 1-15 (2004)
3. Thatcher, T.H., McHugh, N.A., Egan, R.W., Chapman, R.W., Hey, J.A., Turner, C.K., Redonnet, M.R., Seweryniak, K.E., Sime, P.J., Phipps, R.P., Am. J. Physiol. Lung Cell Mol. Physiol. 289(2): L322-328 (2005)
4. D'hulst, A.I., Vermaelen, K.Y., Brusselle, G.G., Joos, G.F., Pauwels, R.A., Eur. Respir. J. 26(2): 204-213 (2005)
5. Lebrun et al. (2006), SOT POSTER
6. Wouters, K., Shiri-Sverdlov, R., van Gorp, P.J., van Bilsen, M., Hofker, M.H., Clin. Chem. Lab. Med. 43(5): 470-479 (2005)
7. International Organization of Standardization. International Standard ISO 3402, 4.th ed., 1999.
8. Haussmann, H.J., Anskeit, E., Becker, D., Kuhl, P., Stinn, W., Teredesai, A., Voncken, P., Walk, R.-A., Toxicol. Sci. 41(1): 100-116 (1998)
9. International Organization of Standardization: International Standard ISO 3308, 4th ed., 2000
10. American Association for Laboratory Animals Science Policy on the Humane Care and Use of Laboratory Animals, Lab. Anim. Sci. 41: 91 (1991)

This work was funded by PHILIP MORRIS USA.
EUROTOX 2006, September 20 to 24, 2006, Dubrovnik, Croatia