Lung Tumor Response in Swiss SWR/J and A/J Mice after Exposure to Cigarette Mainstream Smoke

W. Stinn[1], A. Büttner[1], J.H.E. Arts[2], C.F. Kuper[2], and H.-J. Haussmann[1];
[1]PHILIP MORRIS Research Laboratories GmbH, Cologne, Germany; [2]TNO Quality of Life, Zeist, The Netherlands

Walter.Stinn@pmintl.com

Introduction

The risk of developing lung tumors from cigarette smoking in humans has been well established (IARC, 2004). Despite this causality establishing reproducible and validated animal inhalation models for lung cancer induced by tobacco smoking has proven difficult. These models might be useful for evaluating potential strategies for reducing the harm from smoking.
Here we report on 2 mouse strains as possible models for lung tumorigenicity: A/J and Swiss SWR/J mouse.
A/J mouse studies have shown:

- High spontaneous lung tumor incidence and multiplicity probably due to activation of and/or mutations in the K-ras gene mapped to the pulmonary adenoma susceptibility 1 (Pas1) locus (Lin et al., 1998)
- Reproducible enhancement of lung tumors after exposure to an environmental tobacco smoke surrogate (ETSS) for 5 months followed by 4 months without further exposure (Witschi et al., 2004; Stinn et al., 2005).
- Inconsistent lung tumor response after exposure to cigarette mainstream smoke (Finch et al., 1996; D'Agostini et al., 2001; Curtin et al., 2004)

Swiss SWR mouse studies have shown:

- Low spontaneous lung tumor incidence (<10%) and multiplicity (Witschi et al., 2002; De Flora et al., 2003)
- Sensitive to the induction of lung tumors following exposure to ETSS (Witschi et al., 2002; De Flora et al., 2003)

Objective

- Investigate and compare lung tumor incidence and multiplicity in **A/J mice** and **Swiss SWR/J mice** after chronic inhalation exposure to diluted cigarette mainstream smoke with regard to
 - the **dose dependency** of whole smoke,
 - the influence of a **post-exposure period**, and
 - the influence of **different mainstream smoke aerosol phases**.

Materials and Methods

- Male A/J and Swiss SWR/J mice bred under specific-pathogen-free conditions (Jackson Laboratories, Maine, U.S.A.)
- Whole-body exposure in 2.3-m^3 Hazleton chambers
 - 6 hours/day; 5 days/week; 9 months with dissection after
 - 5 months exposure: control and high whole smoke groups
 - 4 months post-exposure: all groups
- Mainstream smoke (MS) from the Reference Cigarette 2R4F (total particulate matter (TPM) yield: 11.6 mg/cig.) generated under ISO conditions
 - whole smoke (**WS**) at TPM conc. of 120 mg/m^3 (low) and 240 mg/m^3 (high)
 - gas-phase-depleted particle phase (**PP**) generated by passing WS through an activated charcoal filter; TPM conc. of 240 mg/m^3
 - gas phase (**GP**) generated by passing WS through an electrostatic filter; carbon monoxide (CO) conc. equivalent to 240 mg/m^3
- End points
 - body weight (weekly)
 - carboxyhemoglobin (HbCO) in blood
 - gas chromatography of methane after reduction of the CO released from hemoglobin
 - corticosterone in plasma (stress marker)
 - radioimmunoassay
 - inflammation (free lung cells (FLC) in bronchoalveolar lavage fluid (BALF))
 - FLC differentiation in BALF
 - macroscopic lung tumor multiplicity and incidence
 - macroscopic counting of nodules after immersion in Tellyesniczky's fixative for 1 day followed by fixation in ethanol
 - microscopy of lung sections
 - histopathology of hematoxylin/eosin (HE)-stained step serial lung slides (distance: 100 µm)
 - lung tumor classification according to WHO guidelines (2001)
- Statistical tests
 - One-way ANOVA followed by Dunnett: plasma corticosterone and FLC in BALF
 - Chi-square test: lung tumor incidence
 - Kruskal-Wallis followed by Dunnett: lung tumor multiplicity

 Biological data represent means ± SE
 * in figures: statistically significant at p <0.05 compared to fresh air control
- The study was conducted in accordance with the
 - OECD principles of Good Laboratory Practice (1998)
 - American Association for Laboratory Animal Science Policy on the Human Care and Use of Laboratory Animals (1991)

X:\IC\Poster\WST_AACR_2006.vsd

Results

Test Atmospheres

Chemical Analysis

- Target concentrations were met.
- The size of the particles (0.4 to 0.5 µm) indicates that they were respirable for the mice.

Relative to Whole Smoke (WS)

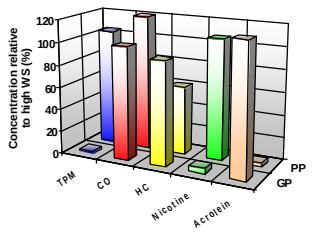
Concentration relative to high WS (%)
TPM
CO
HC
Nicotine
Acrolein
PP
GP

- Gas-phase-depleted particle phase (PP)
 - CO 20% higher
 - hydrocarbons in gas phase (HC) reduced by ~40%
 - acrolein reduced by >95%
- Gas phase (GP)
 - particles and nicotine reduced by more than 94%
 - CO and HC similar to high WS

Body Weight

- Exposure period
 - body weight gain reduction: up to 12% in A/J and 7% in Swiss SWR/J mouse
 - reduction caused by PP more pronounced than by GP in the first 100 days
- Post-exposure period
 - complete recovery

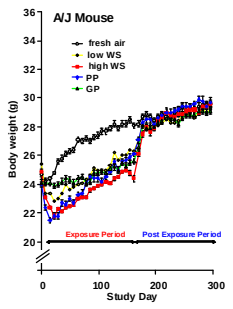
A/J Mouse
fresh air
low WS
high WS
PP
GP
Body weight (g)
Exposure Period
Post Exposure Period
Study Day

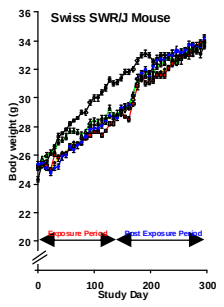
Swiss SWR/J Mouse
Body weight (g)
Exposure Period
Post Exposure Period
Study Day

Plasma Corticosterone Level

- Dose-dependent increase in WS groups
- Similar stress effect in PP and GP groups

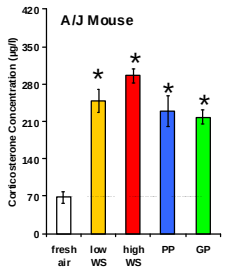
A/J Mouse
Corticosterone Concentration (µg/l)
fresh air
low WS
high WS
PP
GP

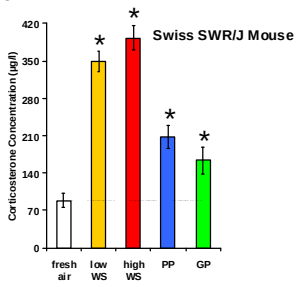
Swiss SWR/J Mouse
Corticosterone Concentration (µg/l)
fresh air
low WS
high WS
PP
GP

determined twice during exposure period; 7 to 8 mice per group

Inflammation

- End of exposure period
 - A severe inflammation was indicated in the high WS group by a severe increase in neutrophils (up to 30% of FLC) and macrophages in BALF.
- End of post-exposure period
 - Both cell types returned nearly to control values.

Neutrophils

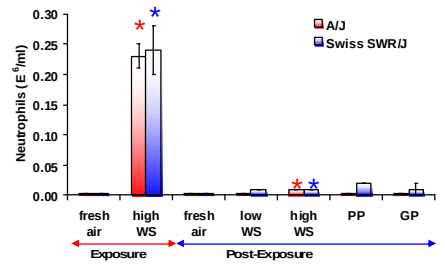
A/J
Swiss SWR/J
Neutrophils (E^6/ml)
fresh air
high WS
fresh air
low WS
high WS
PP
GP
Exposure
Post-Exposure

Alveolar Macrophages

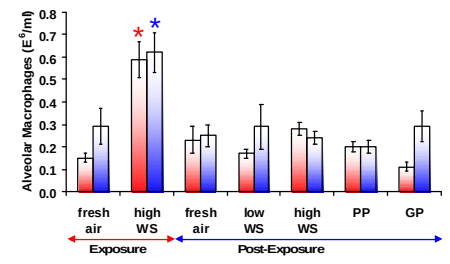
Alveolar Macrophages (E^6/ml)
fresh air
high WS
fresh air
low WS
high WS
PP
GP
Exposure
Post-Exposure

7 to 10 mice per group and time point

Quality Parameters

Mortality

- In both mouse strains, no additional mortality caused by WS, PP, and GP
 - during exposure: up to 5%
 - during post-exposure: 2 to 11%

Steady-State Carboxyhemoglobin (HbCO)

- In both mouse strains the HbCO values were in line with the inhaled CO concentration.
 - low WS groups: 14 to 28%
 - high WS, PP, and GP groups: 23 to 43%

References
American Association for Laboratory Animal Science Policy on the Humane Care and Use of Laboratory Animals, *Lab. Animal Sci.* 41: 91 (1991)
Curtin et al., *Toxicol. Sci.* 81: 26-34 (2004)
D'Agostini et al., *Int. J. Oncol.* 18: 607-615 (2001)
Finch et al., *Cancer Lett.* 99: 161-167(1996)
IARC monographs on the evaluation of carcinogenic risks to humans, International Agency for Research on Cancer, Vol. 83, pp. 1452, 2004
OECD Principles on Good Laboratory Practice (1998), Paris, ENV/MC/CHEM(98)17
Manenti et al., *Cancer Res.* 57: 4164-4166 (1997)
Malkinson et al., *Cancer Res.* 52: 2670-2676 (1992)
Stinn et al., *Inhal. Toxicol.* 17: 549-576 (2005)
WHO, International Classification of Rodent Tumors, The Mouse, 2001
Witschi et al., *Inhal. Toxicol.* 16: 27-32 (2004)
Witschi, *Toxicol. Sci.* 84 (1): 81087 (2005)

Macroscopic Lung Tumor Evaluation

End of exposure period (WS high group compared to control)

- No difference in lung tumor incidence and multiplicity

End of post-exposure period (all groups)

- Enhanced lung tumor incidence and multiplicity
- **WS dose dependency and strain specificity**
 - dose-dependent increase in lung tumor incidence and multiplicity in both strains
 - A/J mouse: up to ~3-fold (multiplicity)
 - Swiss SWR/J mouse: up to ~2-fold (multiplicity)
- Influence of **different mainstream smoke aerosol phases**
 - A/J mouse: similar for PP and WS (incidence and multiplicity)
 - Swiss SWR/J mouse: PP accounted for ~50% of the lung tumor multiplicity in the WS group
 - both mouse strains: GP failed to enhance lung tumor incidence and multiplicity.
 - This was unexpected because in previous studies with mice exposed to ETSS, lung tumor multiplicity and incidence were the same for whole smoke and gas phase (Witschi, 2005).

Lung Tumor Multiplicity

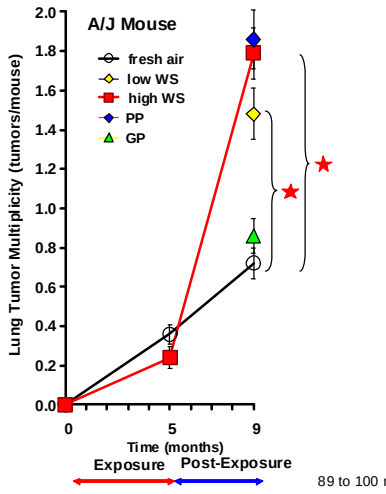
A/J Mouse
fresh air
low WS
high WS
PP
GP
Lung Tumor Multiplicity (tumors/mouse)
Time (months)
Exposure
Post-Exposure

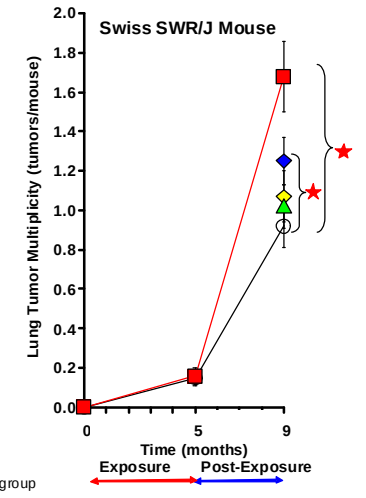
Swiss SWR/J Mouse
Lung Tumor Multiplicity (tumors/mouse)
Time (months)
Exposure
Post-Exposure

89 to 100 mice/group

Lung Tumor Incidence

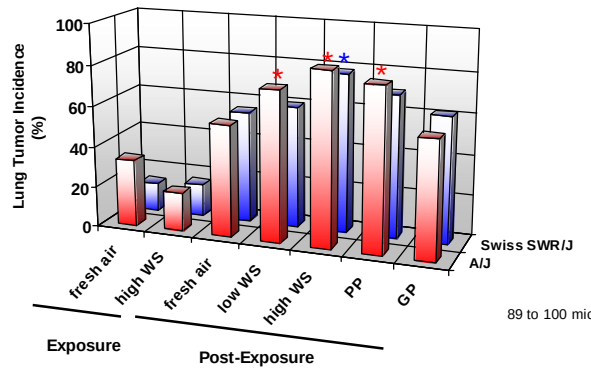
Lung Tumor Incidence (%)
fresh air
high WS
fresh air
low WS
high WS
PP
GP
Swiss SWR/J
A/J
Exposure
Post-Exposure

89 to 100 mice/group

Microscopic Lung Tumor Evaluation

Macroscopic vs Microscopic Results

- Adenomas and carcinomas detected during microscopic examination of lung step serial sectioning confirmed the macroscopic lung tumor evaluation.
- Smoke exposure did not change the lung tumor spectrum.
- Bronchiolo-alveolar adenoma was the most prominent lung tumor type.
 - In humans, carcinomas are the most prominent.

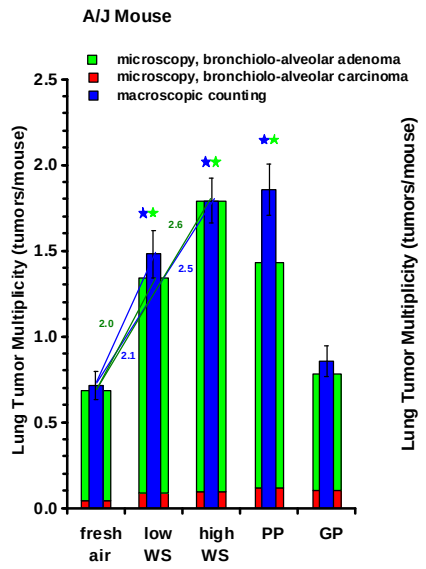
A/J Mouse
microscopy, bronchiolo-alveolar adenoma
microscopy, bronchiolo-alveolar carcinoma
macroscopic counting
Lung Tumor Multiplicity (tumors/mouse)
fresh air
low WS
high WS
PP
GP

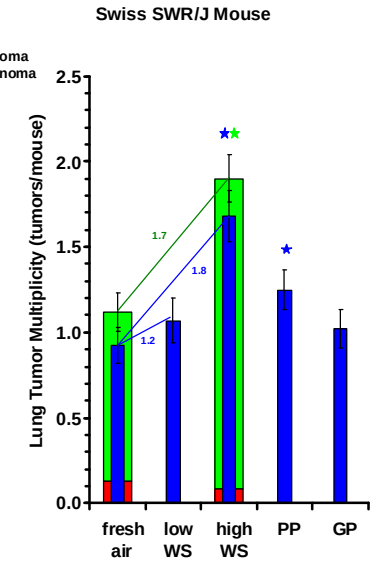
Swiss SWR/J Mouse
Lung Tumor Multiplicity (tumors/mouse)
fresh air
low WS
high WS
PP
GP

9 months; 91 to 97 mice/group

Conclusion

- Lung tumor response after exposure to cigarette mainstream smoke was similar in A/J and Swiss SWR/J mice.
- Lung tumor incidence and multiplicity in A/J and Swiss SWR/J mice increased with increasing whole smoke dose.
- There appears to be a need for a post-exposure period to see an increase in lung tumor incidence and multiplicity in **both mouse strains**.
- In these mouse strains, lung tumor incidence and multiplicity are enhanced primarily by gas-phase-depleted particle phase and not by gas phase.

AACR 2006, April 1 to 5, 2006, Washington, DC, USA
Parts of the A/J mouse results presented at SOT 2005
This work was funded by PHILIP MORRIS USA.