Identification of Compounds of Biological Interest in Tobacco Extracts by Liquid Chromatography Coupled to Mass Spectrometry

Rossella Avallone[1], Andrea Raffaelli[2], Jean-Pierre Schaller[3], Mario Baraldi[1]

1. Dipartimento di Scienze Biomediche, Sezione di Farmacologia, Via Campi 287, 41100 Modena, Italy.
2. CNR-ICCOM, Sezione di Pisa, Dipartimento di Chimica e Chimica Industriale, Via Risorgimento, 35, 56126 Pisa, Italy.
3. Philip Morris International R&D, Applied Research Department, Quai Jeanrenaud 56, 2000 Neuchâtel, Switzerland.

Introduction

A challenging question in benzodiazepine (BDZ) research is the identification of endogenous BDZ ligands in the brain, which could modulate γ-aminobutyric acid neurotransmission. Endogenous substances with benzodiazepine-like activity play a role in different pathologies like hepatic encephalopathy or idiopathic recurring stupor. Several substances with benzodiazepine-like activity have been also found in food and in several officinal plants used in folk medicine as hypnotic or tranquillisers such as *Matricaria chamomilla* or *Ceratonia siliqua*.

The aim of the present work was to determine the possible presence of BDZ-like compounds in leaves of *Nicotiana tabacum* and to test their ability to bind central or peripheral BDZ receptors.

Extraction and Biological Tests

Tobacco leaves were extracted with methanol and the crude extract was chromatographed at 0.8 ml/min on a LiChrospher 100 RP-18 column (250x4.0 mm; 5 μm) equilibrated with 80% water/1% Formic Acid (FA) and 20% acetonitrile. The sample was analysed using a water/1%FA and acetonitrile gradient at 0.5% from 20 to 58% acetonitrile. 75 fractions were collected, lyophilised, and tested for their ability to inhibit [^{3}H]RO 15-1788 specific binding to central BDZ binding sites or [^{3}H]PK 11195 specific binding to peripheral BDZ binding sites (PBR). The crude extract, as well as the separated HPLC fractions, have been analyzed by HPLC-MS and HPLC-MS-MS in order to identify any substances related to the observed biological activity.

LC-MS and LC-MS-MS Analysis

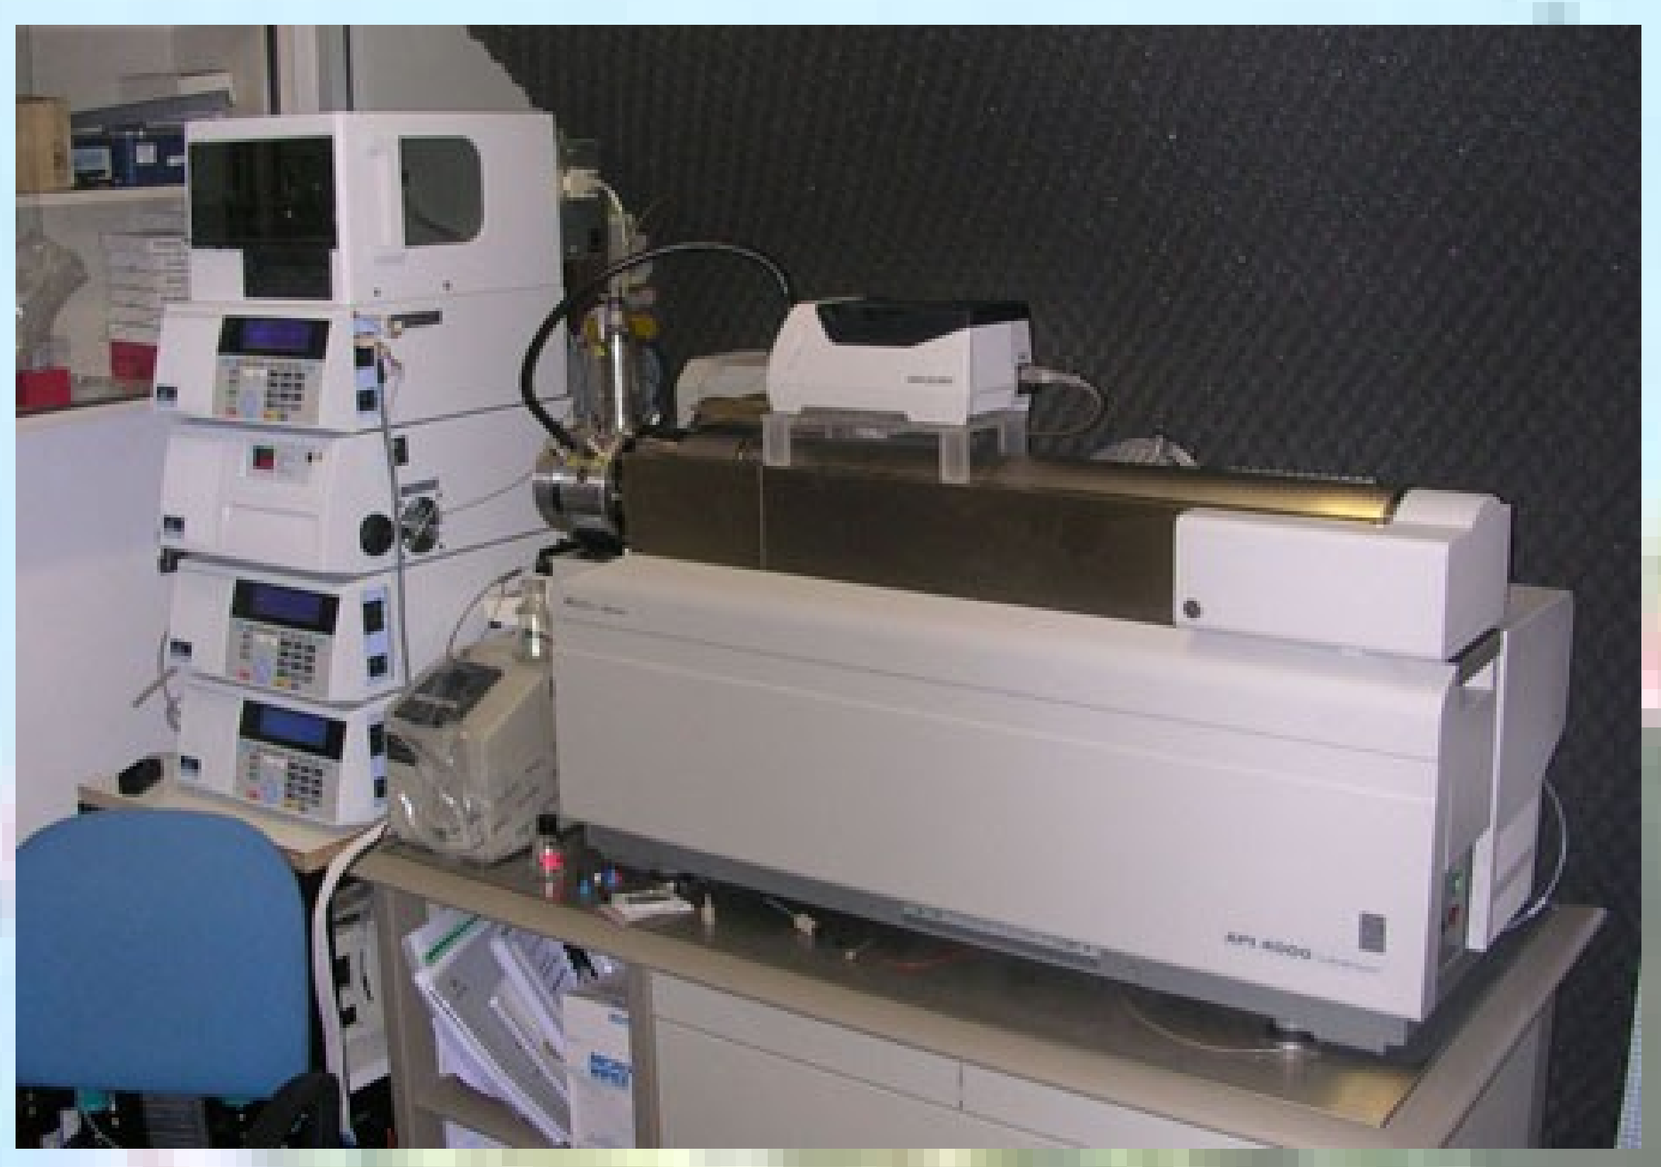

- AB Sciex API 4000 triple quadrupole mass spectrometer
- Perkin Elmer Series 200 Micro binary HPLC system (high pressure mixing)
- Perkin Elmer Series 200 Autosampler
- Perkin Elmer Series 200 Column Oven

- HPLC Column: Phenomenex Gemini C18 (2x50 mm, 5 μ)
- Mobile Phase: Water/Acetonitrile, 0.1% Formic Acid
- Flow Rate: 200 μl/min, linear gradient
- ESI-MS: Scan 100-600 Th in 3 s, Positive and negative ions
- ESI-MS-MS: Product Ions scan of selected precursor, CE 30 eV, Collision Gas Pressure (N_2): 3.4x10^{-5} Torr
- Samples:
 - Crude Tobacco Extract
 - HPLC fractions (1-12)

Cp	MW	Pol.	F1	F2	F3	F4	F5	F6	F7	F8	F9	F10	F11	F12
A	354	Ps	-		-	(+)	+	+	+	-	-	-	-	-
B	370	Ps	-	-		-	-	-	+	++	++	++	+	-
C	304	Ps				-	-	-	-	-	-	-	-	+
D	315	Ps					-	-	-	-	-	-	-	+
E	354	Ng			-	+	++	++	+	(+)		-	-	-
F	416	Ng	-		-		-	+	++	(+)	-		-	-
G	416	Ng	-				-	-	-	-	++	+	+	(+)

Distribution of the compounds found in the HPLC fractions. The highlighted boxes indicate BDZ-like activity both on the central and the peripheral BDZ receptor

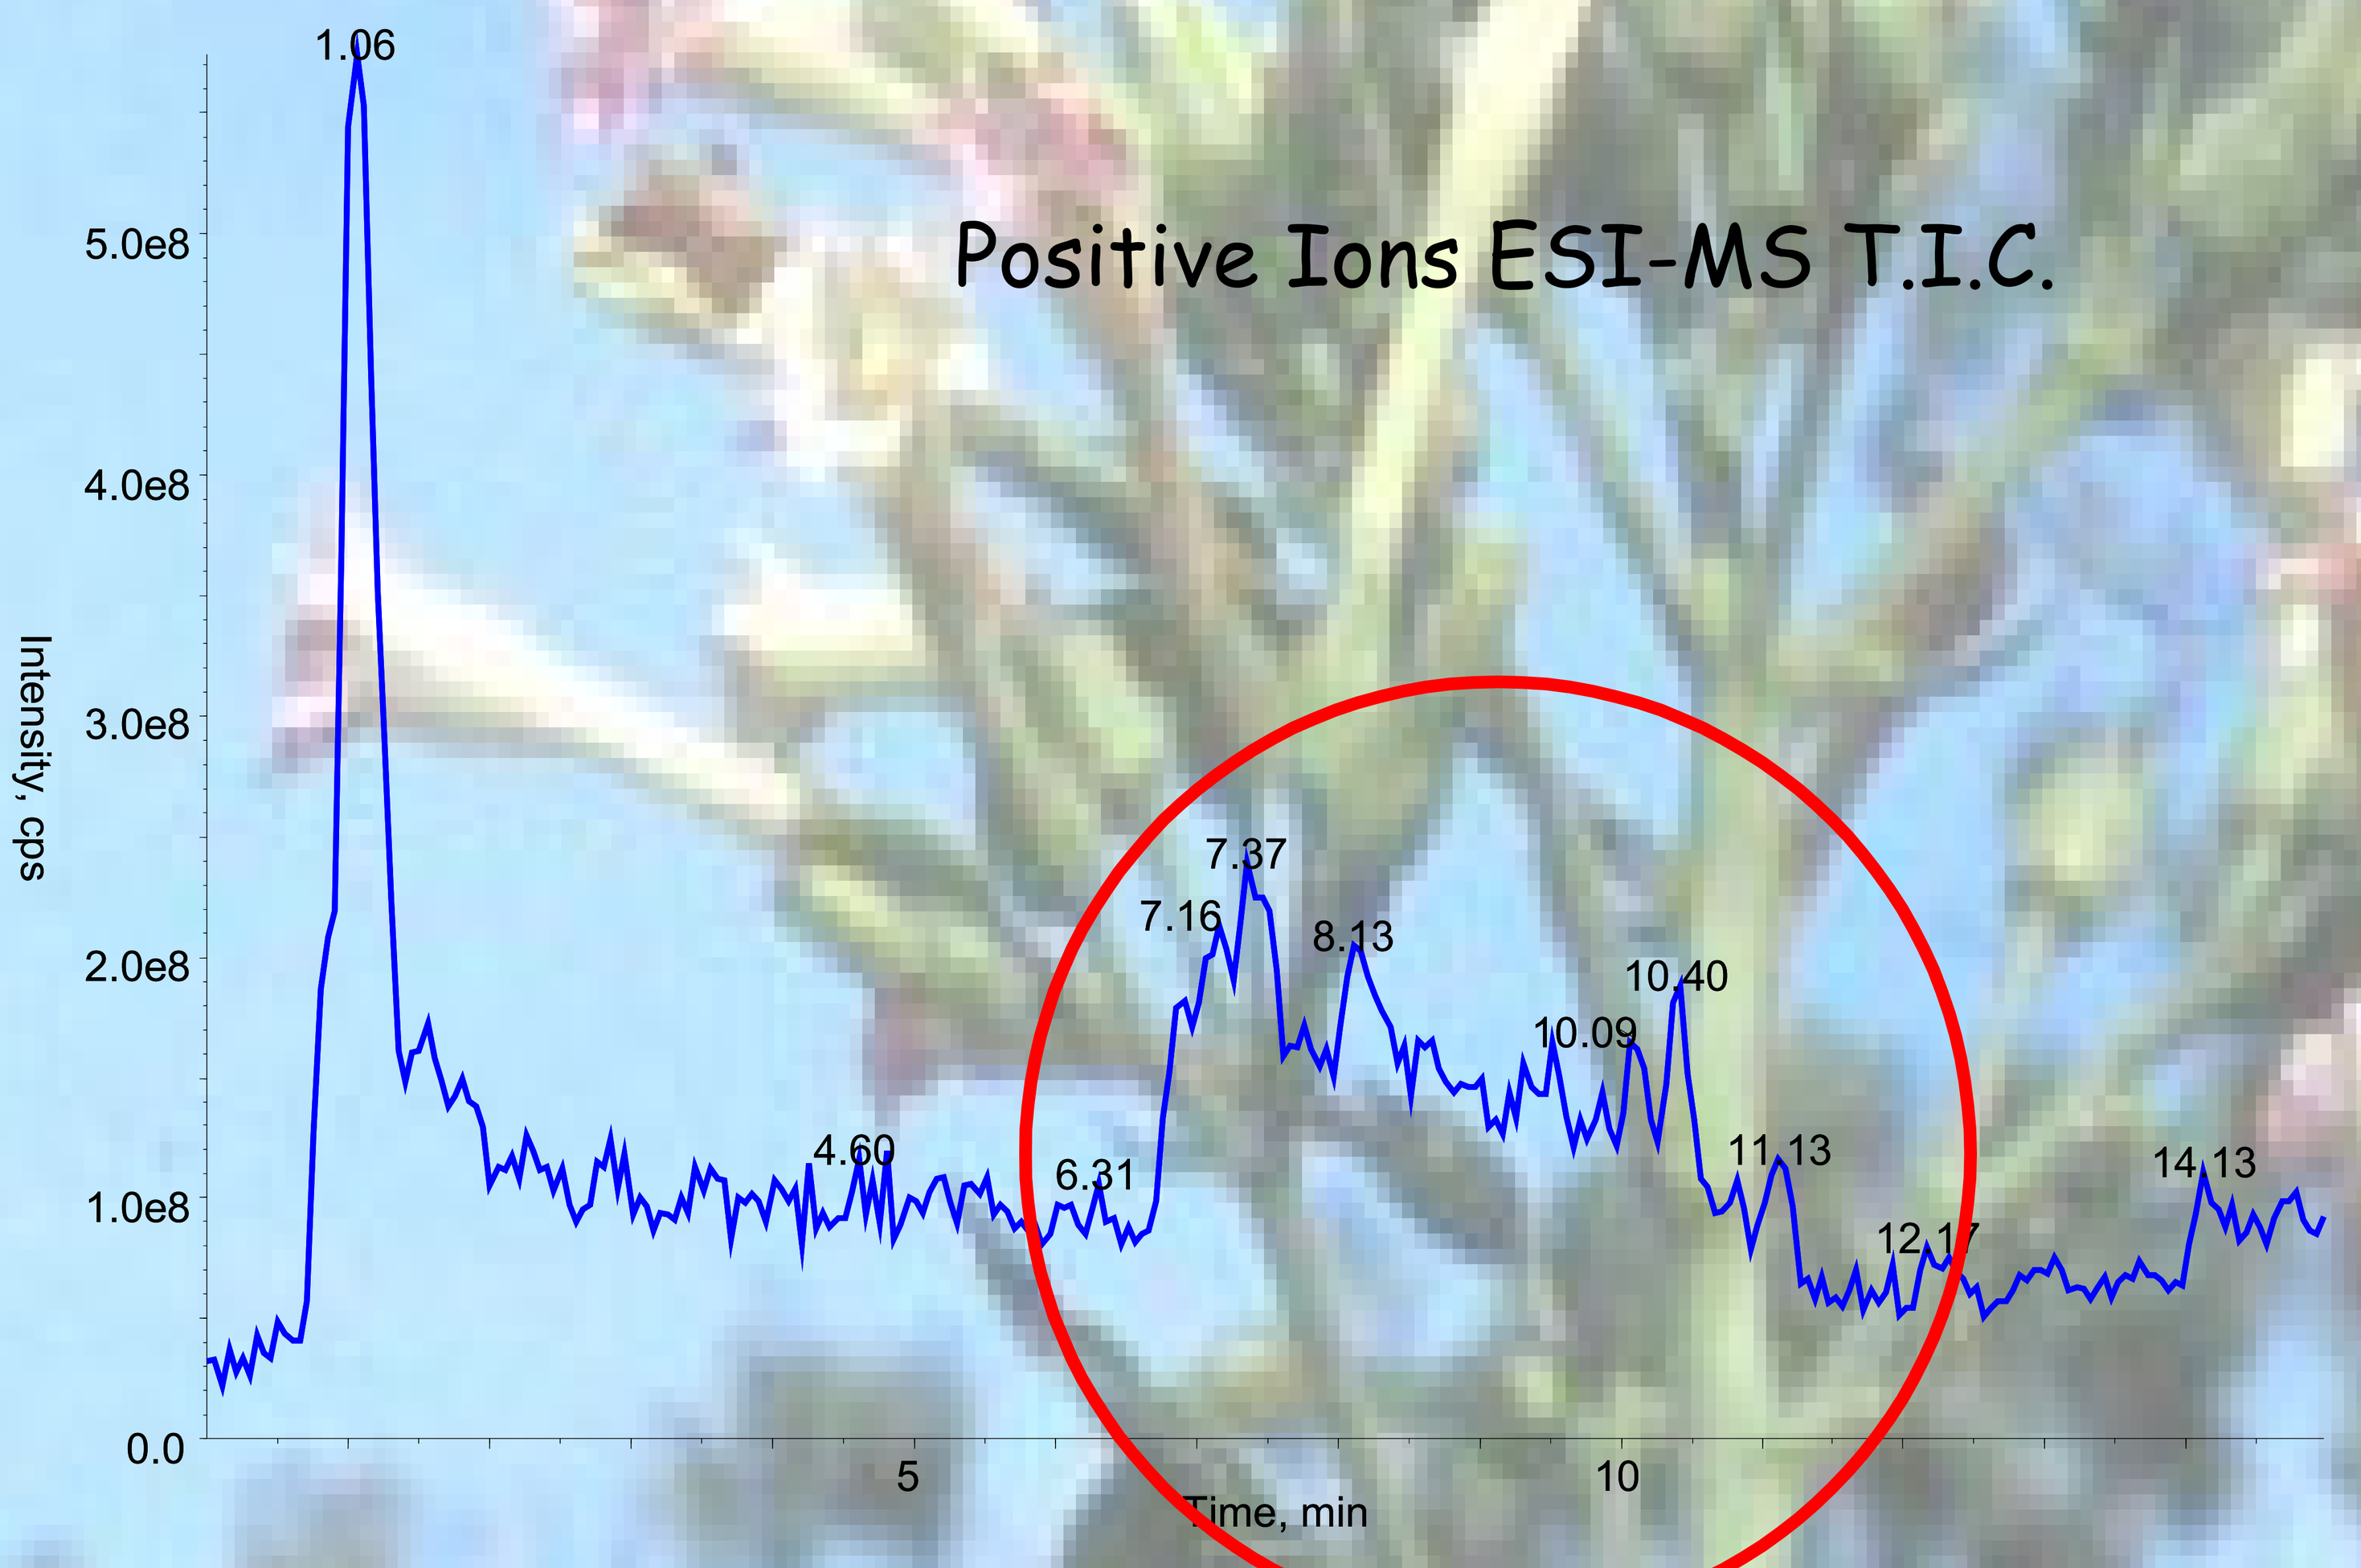
Positive Ions ESI-MS T.I.C.
Intensity, cps
Time, min
1.06
4.60
6.31
7.16
7.37
8.13
10.09
10.40
11.13
12.17
14.13

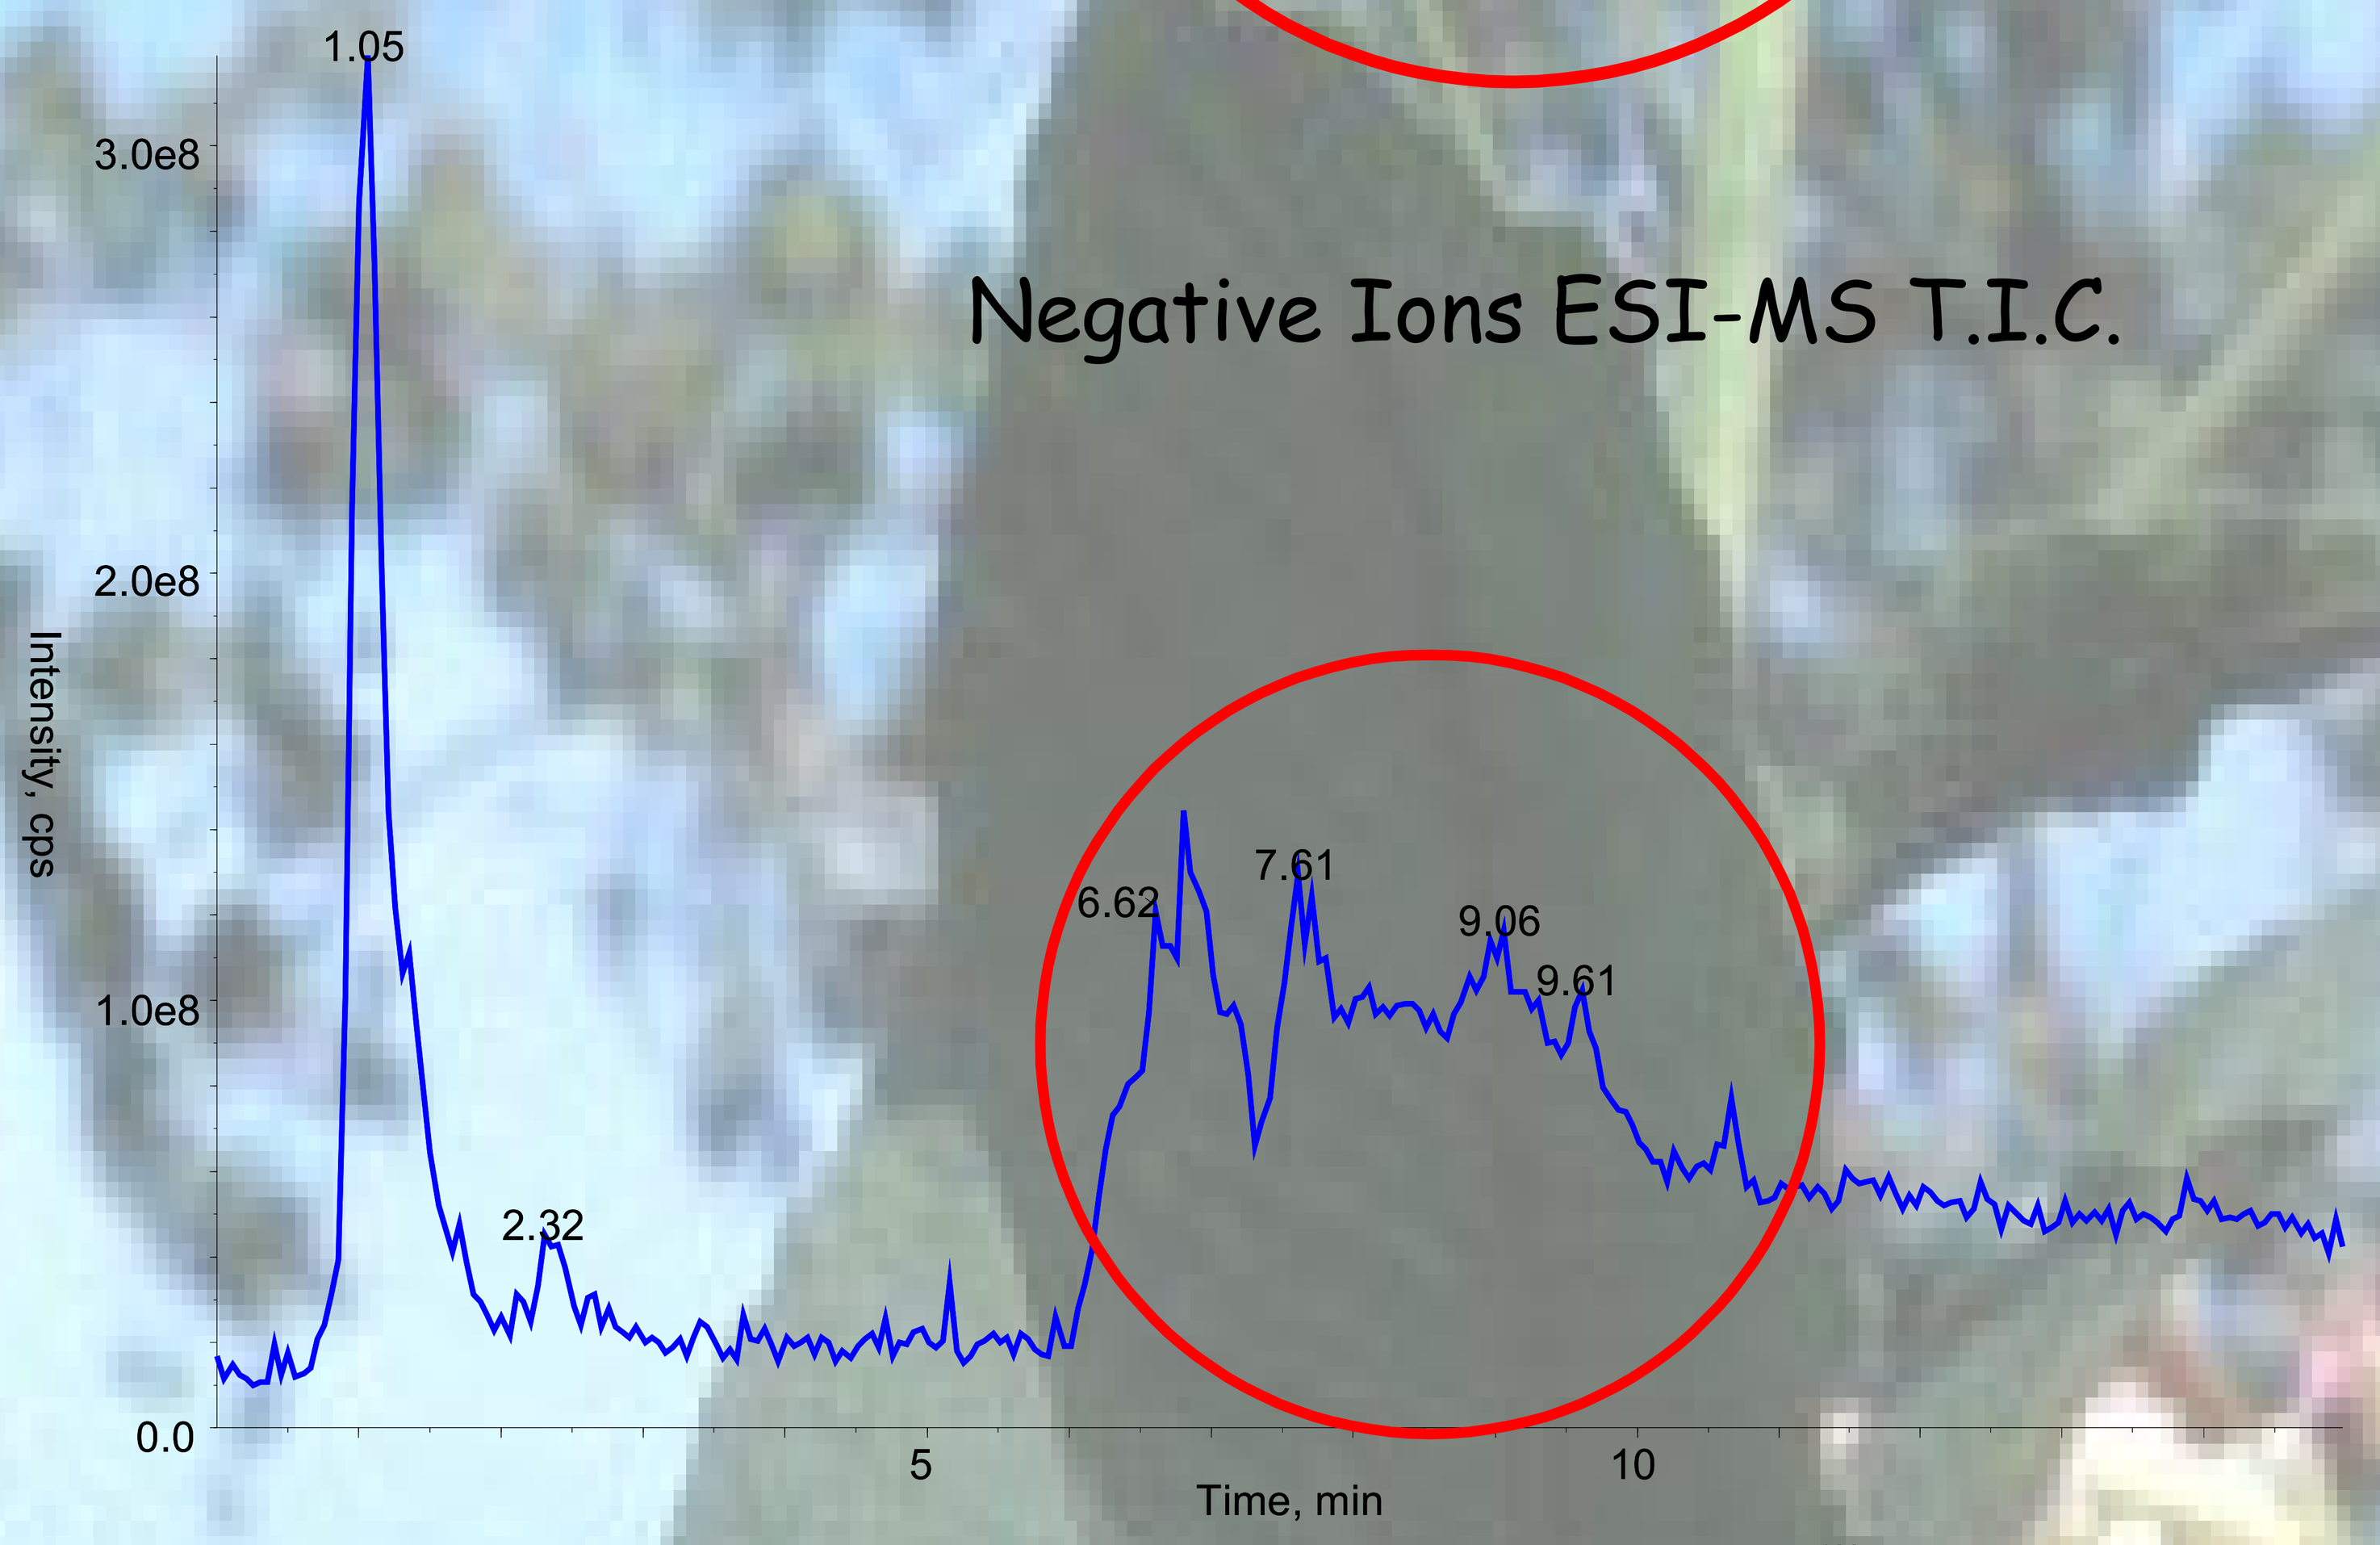
Negative Ions ESI-MS T.I.C.
Intensity, cps
Time, min
1.05
2.32
6.62
7.61
9.06
9.61

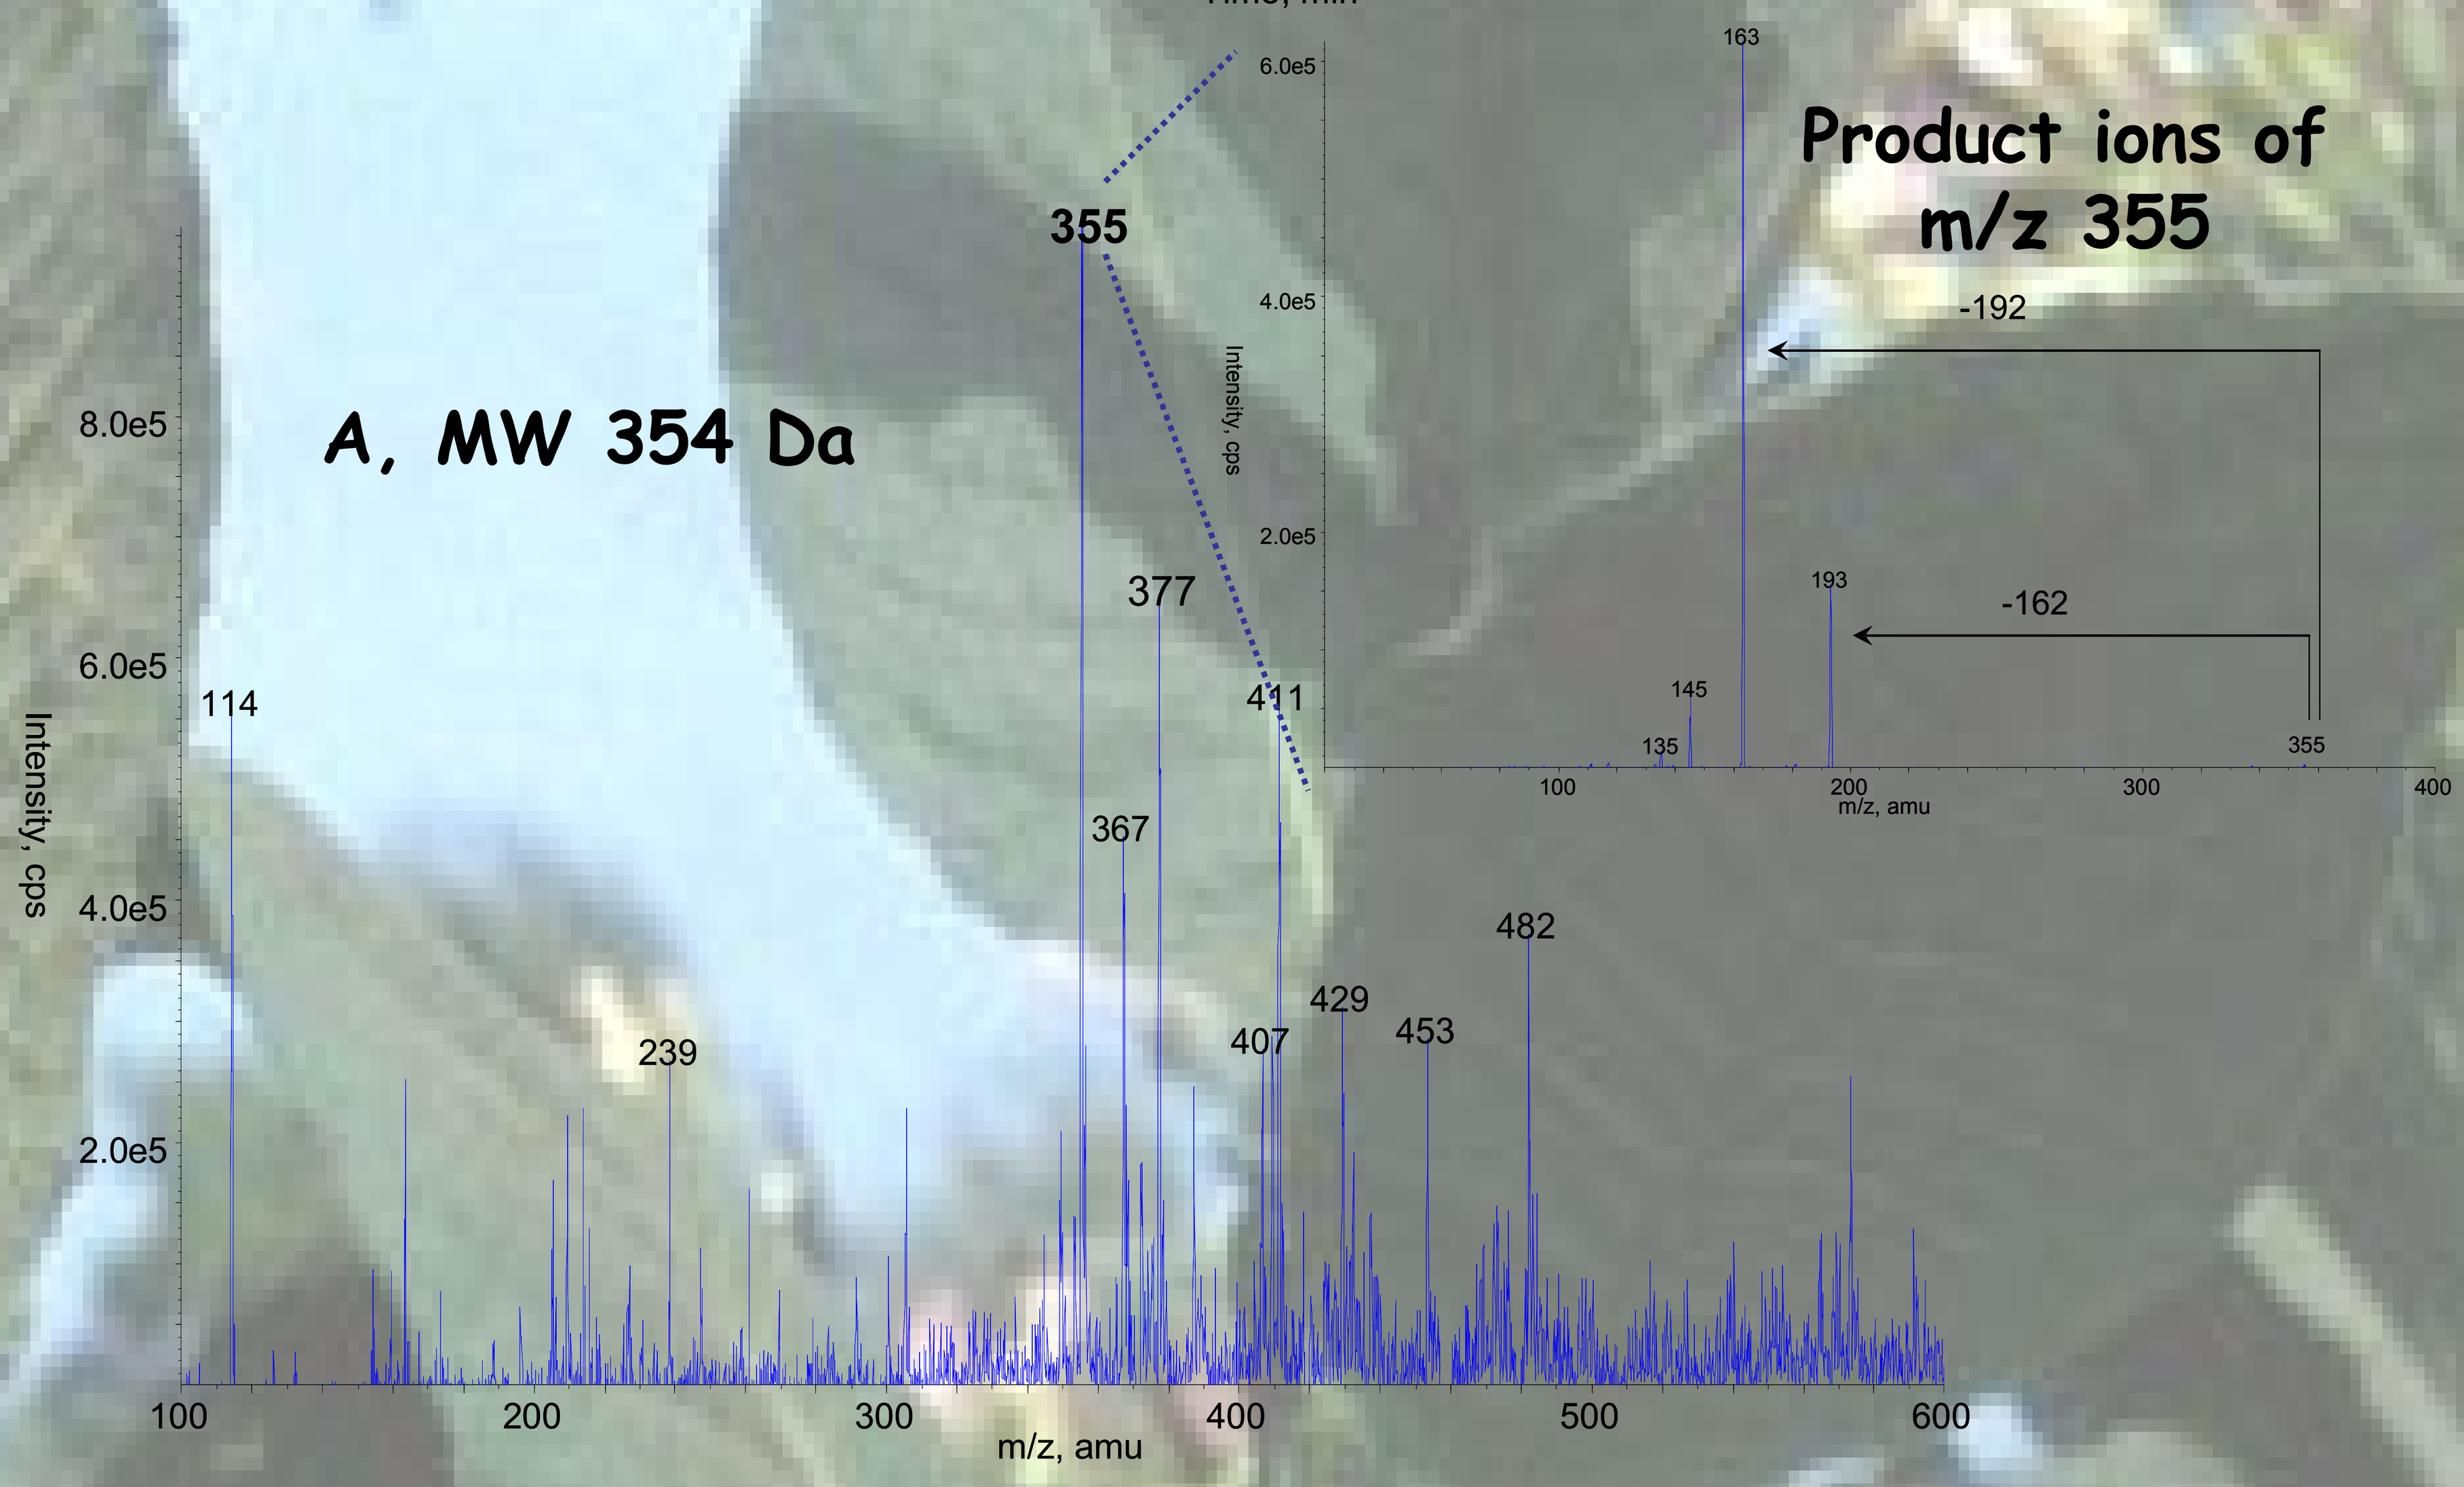
A, MW 354 Da
Product ions of m/z 355
Intensity, cps
m/z, amu
355
114
239
367
377
407
411
429
453
482
163
193
145
135
-192
-162

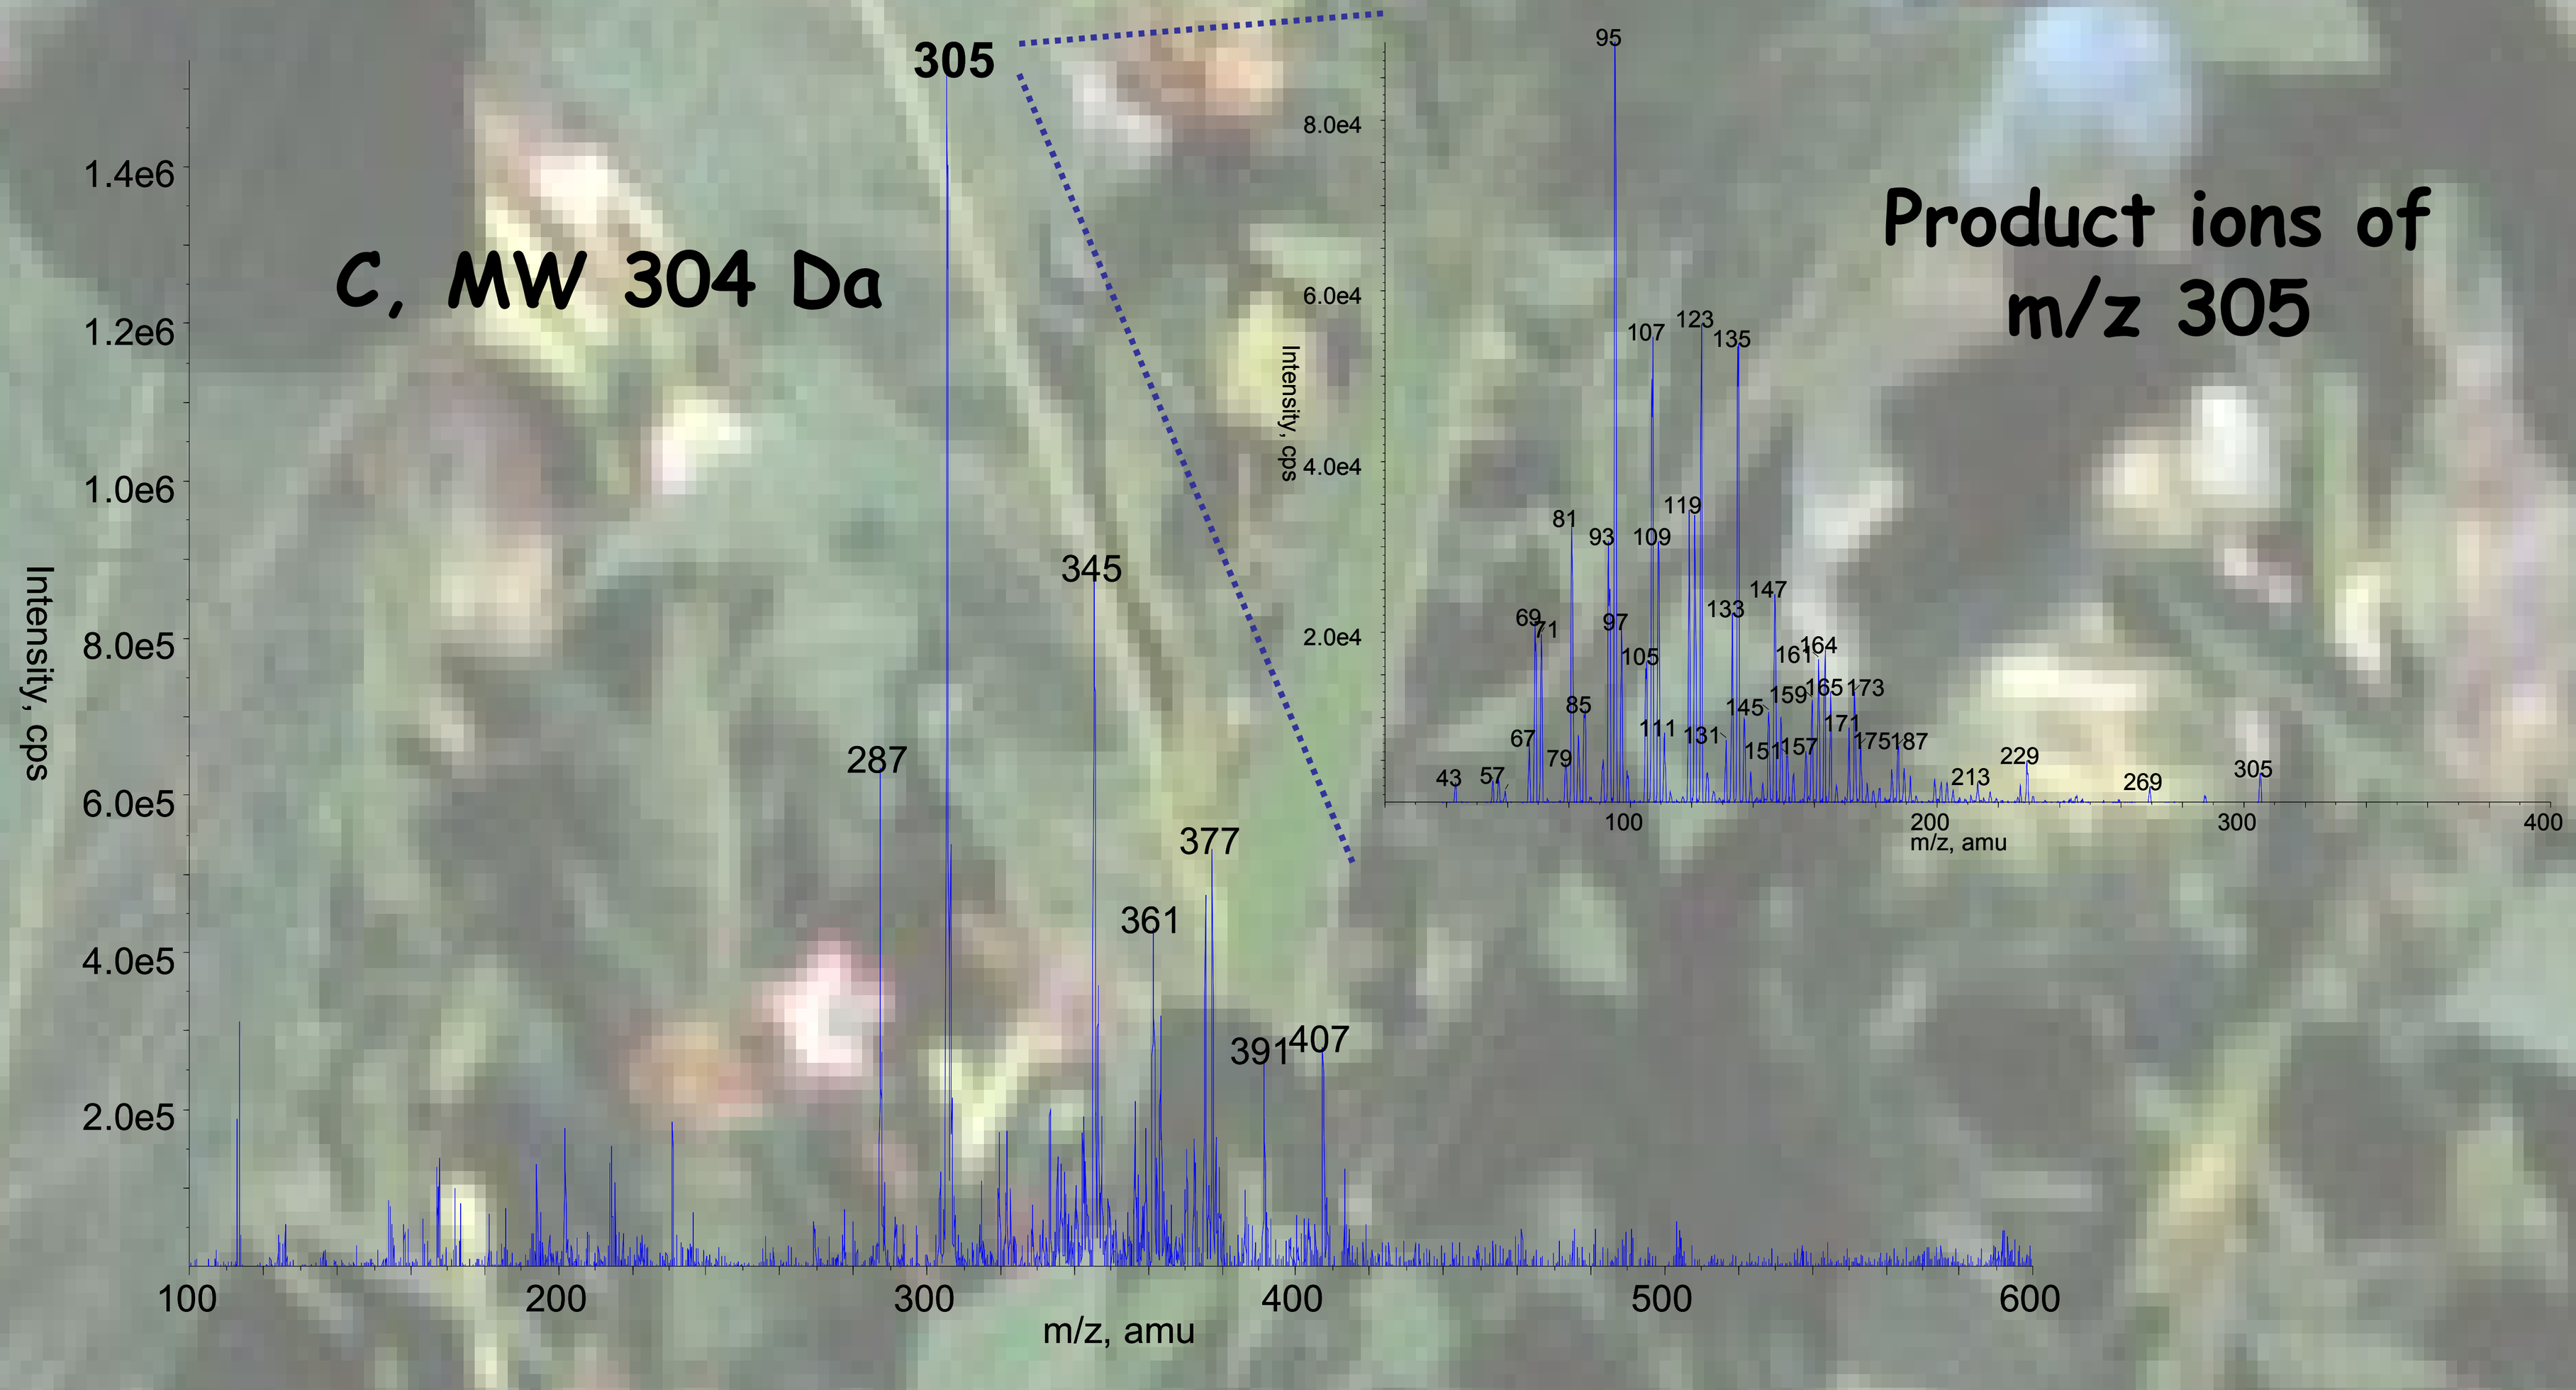
C, MW 304 Da
Product ions of m/z 305
Intensity, cps
m/z, amu
305
287
345
361
377
391
407
95
107
123
135
119
81
93
109

Conclusions

Fractions of tobacco extracts showed BDZ-like activity on the central and/or the peripheral BDZ receptors.

HPLC/MS-MS methods based on Parent and Neutral Loss scans were developed to identify the chemical structure of the biologically active compounds.

Further HPLC-MS-MS and biological activity data crossing is needed to finalize the structure identification.