Towards A Mouse Inhalation Model for Cigarette-Smoke-Induced Lung Tumorigenesis

Walter Stinn[1], Ansgar Buettner[1], Baerbel Friedrichs[1], Sonja Luetjen[1], Stephan Gebel[1], Rosemarie B. Lichtner[2], Frans van Overveld[3*], and Hans-Juergen Haussmann[4]

[1]Philip Morris Research Laboratories, Cologne, Germany; [2]Philip Morris International R&D, Neuchâtel, Switzerland; [3]Philip Morris Research Laboratories, Leuven, Belgium; [4]Toxicology Consultant, Roesrath, Germany.

PMI RESEARCH & DEVELOPMENT

Introduction

The smoking-related risk for lung cancer and other serious diseases is well established (US DHHS, 2004). However, established laboratory animal models for these diseases are not readily available (Schleef et al., 2006; Coggins, 2007). Such models are needed to unravel mechanistic and etiologic aspects of the disease, to investigate chemopreventive and early diagnostic means, and to evaluate risks from novel tobacco products (Haussmann, 2007). Some positive results in rodent lung cancer models with cigarette mainstream smoke (MS) inhalation were recently reported, but have not been reproduced to date (Mauderly et al., 2004; Hutt et al., 2005; Balansky et al., 2007).

Increases in lung tumor response have reproducibly been shown, using a model with the following characteristics (De Flora et al., 2003; Witschi, 2005a; Stinn et al., 2005a):

- 5- to 6-month inhalation exposure to an environmental tobacco smoke surrogate (ETSS)
- A required 4-month post-exposure period to allow the tumorigenic response to be seen
- Mouse strains known for their genetic susceptibility for developing lung adenocarcinoma
 - mainly A/J, but also SWR/J
- Similar tumor response with ETSS whole smoke (WS) or its vapor phase (VP)

However, the majority of lung cancer cases are associated with active smoking, i.e., exposure to MS. Results of MS inhalation studies with this particular study design were not reproducible (Finch et al., 1996; D'Agostini et al., 2001; Curtin et al., 2004; Hamm et al., 2007). This is considered to be due mainly to the variable and sub-optimal study designs employed.

Goal

Our goal is to establish a murine lung tumorigenesis model with cigarette mainstream smoke inhalation.

This model should be biologically relevant, reproducible, and work without the need for a post-exposure period.

Approach

Study 1: Compare results for previous ETSS inhalation studies to MS inhalation
- Susceptible mouse strains (male A/J and SWR/J)
- 5-month exposure + 4-month post-exposure periods
- Whole smoke (WS), particulate phase (PP), and vapor phase (VP)
- Dose dependence (0, 120, and 240 mg TPM/m³ ;sham, low WS, and high WS, respectively)

Study 2: Extend the MS exposure periods (± post-exposure) to long-term
- Up to 18 months of exposure/post-exposure combinations
- Evaluation of the need for a post-exposure period
- Male A/J mice (18 to 36 mice per group) exposed to 0, 150, and 300 mg TPM/m³ (sham, MS-150, and MS-300, respectively)
- Inclusion of mechanistic endpoints

Results, Study 1

Analytical characterization of test atmospheres:

- Efficient separation of PP and VP

Smoke Constituent	Unit of Measure	2R4F MS low WS	high WS	PP	VP
Analyses during the inhalation period:					
TPM	mg/m³	127 ± 11	235 ± 19	235 ± 30	4 ± 11
Nicotine	mg/m³	2.7 ± 0.6	6.2 ± 1.3	6.2 ± 0.8	0.3 ± 0.2
Carbon monoxide	ppm	201 ± 24	322 ± 37	385 ± 56	321 ± 38
Formaldehyde	mg/m³	0.08 ± 0.01	0.12 ± 0.02	0.10 ± 0.02	0.16 ± 0.05
Acetaldehyde	mg/m³	13.5 ± 1.0	24.0 ± 2.0	12.1 ± 3.5	23.3 ± 3.3
Acrolein	mg/m³	0.61 ± 0.14	1.07 ± 0.30	0.03 ± 0.03	1.00 ± 0.28
Offline analyses after termination of the inhalation[a]:					
1,3-Butadiene	mg/m³		0.061 ± 0.047[b]	0.0283 ± 0.013	0.193 ± 0.132
Isoprene	mg/m³		2.4 ± 2.3	0.4724 ± 0.3543	4.9 ± 1.8
Benzene	mg/m³		1.10 ± 0.10	0.1653 ± 0.0118	0.93 ± 0.04
Toluene	mg/m³		2.25 ± 0.35	0.3543 ± 0.0472	1.73 ± 0.04
Acrylonitrile	mg/m³		0.41 ± 0.12	0.0945 ± 0.0118	0.48 ± 0.06
Benz[a]anthracene	µg/m³		0.414 ± 0.027	0.4665 ± 0.0177	0.003 ± 0.002
Benzo[a]pyrene	µg/m³		0.234 ± 0.014	0.261 ± 0.0083	0.002 ± 0.001
Benzo[b]fluoranthene	µg/m³		0.173 ± 0.011	0.196 ± 0.0024	0.001 ± 0.001
Benzo[k]fluoranthene	µg/m³		0.065 ± 0.004	0.072 ± 0.0012	n.d.
Benzo[j]fluoranthene	µg/m³		0.105 ± 0.007	0.1205 ± 0.0035	n.d.
Pyrene	µg/m³		1.17 ± 0.07	1.2519 ± 0.0236	0.01 ± 0.01
Dibenzo[a,e]pyrene	µg/m³		0.009 ± 0.001	0.0106 ± 0	n.d.
Indeno[1,2,3-c,d]pyrene	µg/m³		0.100 ± 0.006	0.1122 ± 0.0035	0.001 ± 0.001
N-Nitrosonornicotine	µg/m³		4.15 ± 0.17	4.4878 ± 0.0709	0.02 ± 0.02
4-(N-Nitrosomethylamino)-1-(3-pyridyl)-1-butanone	µg/m³		4.09 ± 0.15	4.5587 ± 0.0472	0.06 ± 0.05
Phenol	µg/m³		29 ± 8	15.353 ± 4.724	3 ± 2
Catechol	µg/m³		880 ± 7	838.51 ± 15.353	4 ± 2
Hydroquinone	µg/m³		921 ± 26	1008.6 ± 12.991	2 ± 1

[a] PP concentrations corrected by 1.18-fold for loss by carbon filtration; [b] Data probably incorrect (too low).

Body weight development:

- Marked body weight effect during MS exposure (WS ≈ PP > VP)
- Recovery of body weight effects during the post-exposure period

Macroscopic evaluation of lungs (nodule multiplicity):

- No effect at end of exposure period
- Dose-dependent increase in multiplicity at end of post-exposure period
- Activity of WS largely associated with PP; no activity in VP
- Similar responses for both strains

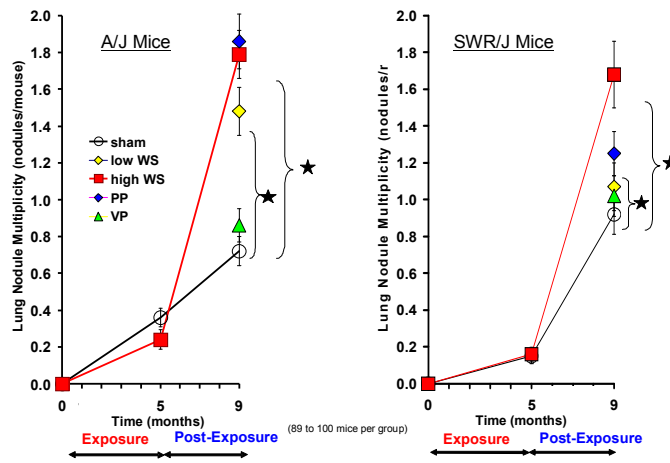
A/J Mice
SWR/J Mice
Lung Nodule Multiplicity (nodules/mouse)
sham
low WS
high WS
PP
VP
Time (months)
Exposure
Post-Exposure
(89 to 100 mice per group)

Microscopic evaluation of lungs (multiplicity of proliferative lesions):

(end of post-inhalation period)

- Increase in bronchioloalveolar adenoma multiplicity
- Similar responses for both strains, good correlation to macroscopic results

Results, Study 2

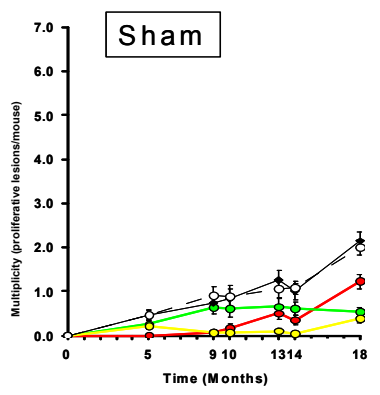
Sham
Multiplicity (proliferative lesions/mouse)
Time (Months)

5 Months Exposure + 13 Months Post-Exposure

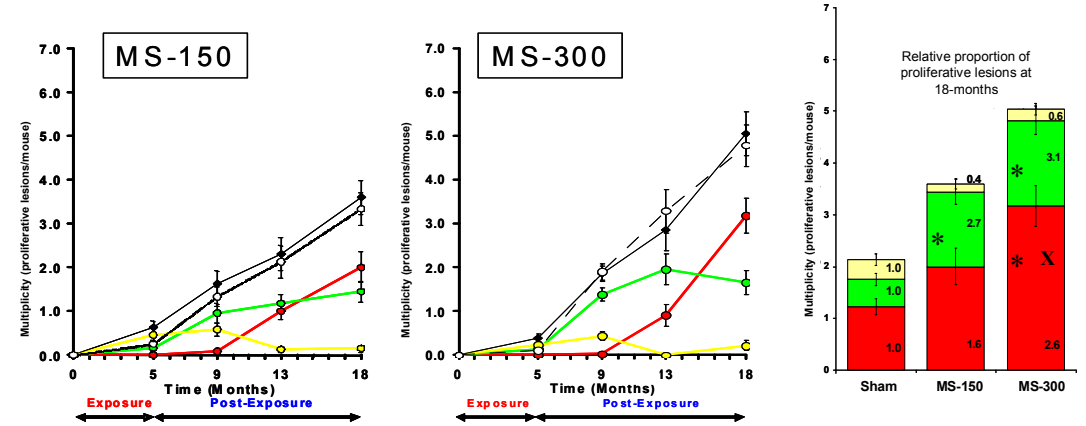
MS-150
MS-300
Time (Months)
Exposure
Post-Exposure
Relative proportion of proliferative lesions at 18-months
Sham
MS-150
MS-300

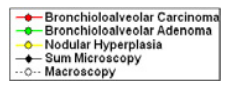
Bronchioloalveolar Carcinoma
Bronchioloalveolar Adenoma
Nodular Hyperplasia
Sum Microscopy
Macroscopy

10 Months Exposure + 8 Months Post-Exposure

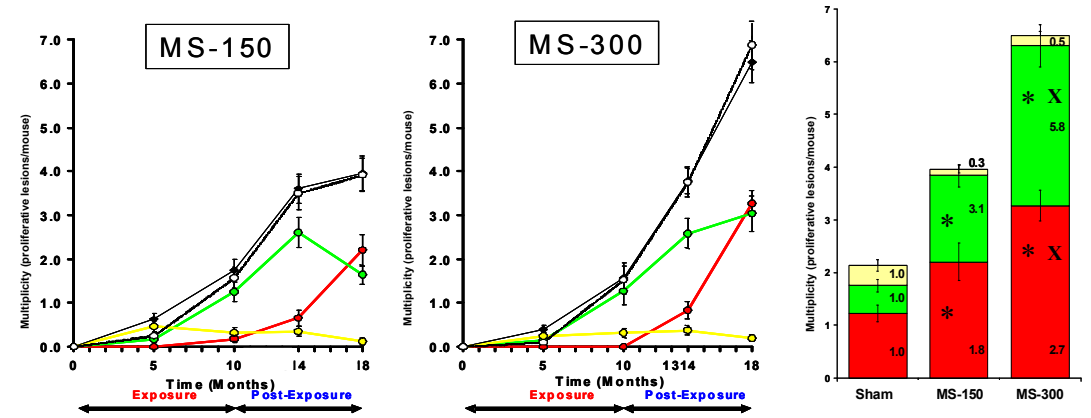
MS-150
MS-300
Time (Months)
Exposure
Post-Exposure
Sham
MS-150
MS-300

18 Months Exposure

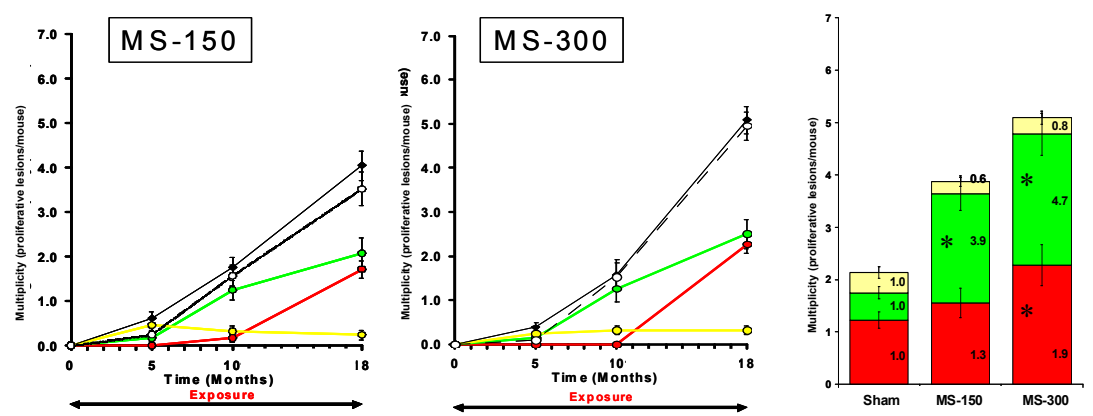
MS-150
MS-300
Time (Months)
Exposure
Sham
MS-150
MS-300

* = statistically significant compared to sham
X = statistically significant compared to MS-150

Microscopic evaluation of lungs (multiplicity of proliferative lesions):

- Increase in spontaneous and MS exposure-induced multiplicity with time (age)
- MS dose-dependent increase in bronchioloalveolar adenomas and adenocarcinomas
- Transient effect on nodular hyperplasia, consistent with being an intermediary step in tumorigenesis
- Tumorigenic potential of long-term MS exposure apparent without the need for a post-exposure period
- Attenuating activity of MS exposure on tumor response and progression still apparent, although fading with increasing exposure period + in parallel to fading stress (corticosterone)
- Good correlation of macroscopic and microscopic results

Corticosterone concentration in blood:

- Trend towards adaptation to the MS exposure-induced stress during long-term exposure

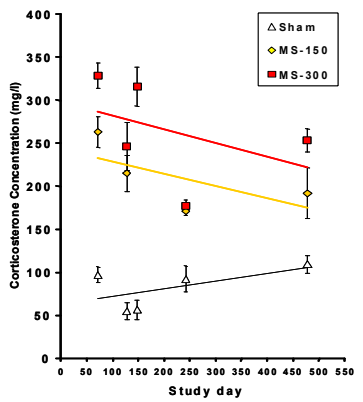
Sham
MS-150
MS-300
Corticosterone Concentration (mg/l)
Study day

Gene Expression (whole lung homogenates):

- Two types of regulation:
 - Up-regulation during exposure, quick recovery during post-exposure:
 - Xenobiotics metabolizing enzymes (Phase 1 and 2)
 - Oxidative stress-responsive enzymes
 - Sustained up-regulation, partial or no recovery during post-exposure:
 - Inflammatory markers (chemokines, proteases, acute phase response)

Exposure / Post-Exposure

gene symbol	0.5 month	2 months	5 months	5 months + 2 days	5 months + 4 months	5 months + 13 months	18 months	
ftl2	1.5	2.1	2.0	1.3	1.4	—	1.9	Oxidative Stress
gclc	6.4	6.7	5.7	1.1	—	—	4.0	
gclm	3.4	3.7	4.1	-1.2	—	-1.4	2.8	
gpx2	2.8	3.1	2.5	-1.4	—	—	—	
gsr	2.5	2.5	2.1	-1.0	-1.3	—	2.0	
hmox1	2.7	4.5	4.4	1.6	—	1.5	4.8	
maff	2.6	2	2.7	-1.2	—	—	2.0	
nqo1	6.4	6.5	6.0	-1.2	—	—	9.4	
txnrd1	4.1	3.6	3.6	1.1	—	—	2.4	
adh7	6.4	10.6	10.0	-1.1	—	—	3.2	Metabolism
aldh3A1	5.6	7.2	7.3	-2.2	—	—	11.0	
akr1B8	4.2	4.3	4.4	1.6	—	—	3.1	
cyp1A1	64.7	67	67.4	-5.1	—	—	100.0	
cyp1B1	10	10.6	8.3	1.5	—	—	44.0	
gsta1	5	6.3	5.3	-1.7	—	—	2.1	
gsta2	5.7	7.2	6.5	-1.7	—	—	2.3	
lox	4.5	3.7	3.7	-1.6	—	—	2.5	
ccl2	2.5	3.2	3.5	4.2	5.2	2.9	5.2	Inflammation
ccl3	6.7	7.3	7.8	8.3	8.9	3.3	9.7	
ccl6	3.8	6.9	6.1	5.7	3.2	2.2	6.1	
ccl20	7.6	5.1	3.6	2.7	3.4	2.1	3.6	
ccl5	-1.9	-2.0	-2.7	-1.6	—	2.2	—	
cxcl1	7.8	9.1	5.6	6.2	3.8	2.5	9.6	
cxcl5	64	59.5	30.1	7.2	2.1	16.7	76.7	
cxcl9	2.1	3.8	4.4	4.9	—	3.7	3.5	
cxcl10	2.3	1.9	3.0	5.3	—	2.2	2.5	
saa3	16.4	17.2	13.6	15.9	10.5	—	28.9	
orm2	3.2	2.8	2.8	2.4	4.4	2.8	33.7	
cd68	2.8	4.7	5.1	4.0	3.3	1.8	4.8	
msr	3.6	10.3	8.4	5.6	3.4	—	6.8	
mmp12	7.1	10.2	10.5	8.8	20.4	2.4	7.1	
timp1	3.4	2.9	2.5	2.0	1.8	—	6.9	
slpi	1.2	3.1	2.6	2.0	2.6	2.8	5.7	
ctsk	5.3	8.9	8.8	5.2	4.3	1.4	6.4	
ctss	2	5.4	5.9	5.3	2.3	—	2.5	

≥ 2-fold increase ≥ 2-fold decrease

Inflammatory processes

(BAL analysis, only sham and MS-300 shown):

- Neutrophils: increase during exposure; recovery during post-exposure
- Lymphocytes: increase during exposure and post-exposure
- Macrophages: no MS-related effect, eosinophils always <1%
- Specific chemokines: increase during exposure and post-exposure (e.g., KC, MCP-1)
- Specific cytokines: increase during exposure; recovery during post-exposure (e.g., IL1a, IFN-g, TNF-a); only marginal MS effect (e.g., IL-1b, IL-13, GM-CSF)
- MMP-9: increase during exposure; recovery during post-exposure

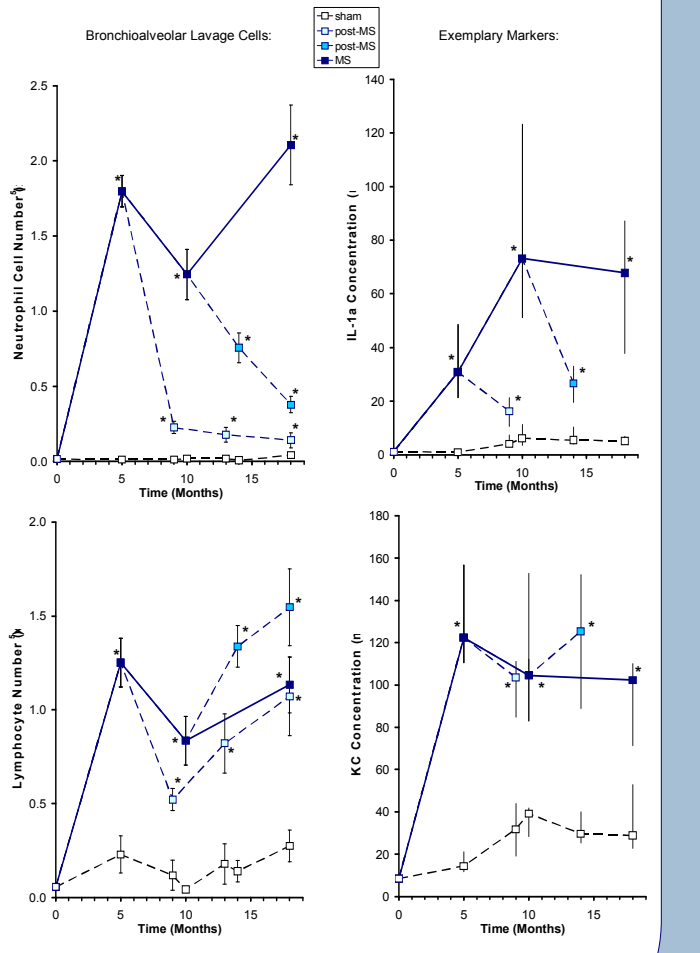
Bronchioalveolar Lavage Cells:
Exemplary Markers:
sham
post-MS
post-MS
MS
Neutrophil Cell Number
IL-1a Concentration
Lymphocyte Number
KC Concentration
Time (Months)

Kras Mutation Pattern (in tumor nodules):

- No significant change in mutation pattern with MS inhalation

Summary

- Mainstream cigarette smoke inhalation (whole-body, 5-month exposure + 4-month post-exposure) dose-dependently and reproducibly enhanced lung tumorigenesis in susceptible mice (A/J and SWR/J mice), provided a study design is being used with the appropriate statistical power (group size, step-serial histopathological examination of lungs).
- The lung tumor response was similar for whole smoke and the particulate phase fraction, with no significant activity found in the vapor phase.
- This phase-specificity is in contrast to the results found with an environmental tobacco smoke surrogate (Witschi, 2005b). A comparative analysis of the test atmospheres (MS, ETSS), which showed an increase in tumorigenesis, did not pinpoint any smoke constituents that might explain the increase in this murine model.
- With prolonged inhalation (18 months), a dose-dependent increase in tumorigenesis was found, even without a post-exposure period, corresponding to an adaptation of the corticosterone response. Still, the tumorigenic process seemed to be somehow restrained with continuous smoke exposure compared to shorter inhalation, which is followed by a post-exposure period (e.g., 10 months exposure + 8 months post-exposure).
- This combination of exposure and post-exposure periods was accompanied by distinct patterns in gene expression and inflammatory responses, such as the sustained up-regulation of inflammatory markers. Their involvement in the tumorigenic response remains to be established. No shift in the mutation pattern in the K*ras* gene was detected.

Overview of MS inhalation studies with A/J mice:

- Statistically significant responses (green) with adequate study design (statistical power and/or step-serial microscopic examination; long duration)
- Higher discriminatory power with long-term exposure (18 months)

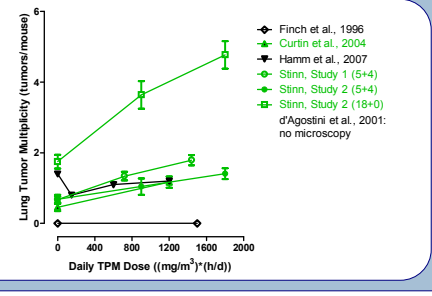
Lung Tumor Multiplicity (tumors/mouse)
Daily TPM Dose ((mg/m³)*(h/d))
Finch et al., 1996
Curtin et al., 2004
Hamm et al., 2007
Stinn, Study 1 (5+4)
Stinn, Study 2 (5+4)
Stinn, Study 2 (18+0)
d'Agostini et al., 2001: no microscopy

Conclusions

- **A reproducible and dose-dependent long-term mainstream cigarette smoke inhalation model for pulmonary adenoma and adenocarcinoma seems to be feasible using A/J mice—provided an adequate study design is used.**
- **The final study design of this model needs to consider that smoke inhalation seems to affect both accelerating and decelerating tumorigenic processes. A post-exposure period is not required with long-term exposure.**
- **Although mechanistic studies are already available, more information is needed to appreciate the relevance of this model for the human disease. However, success against this objective is limited due to the limited understanding of the etiology and pathogenesis of the human disease.**

Hypothesis

Balance of accelerating and decelerating tumorigenic processes in the A/J mouse model for pulmonary adenocarcinoma

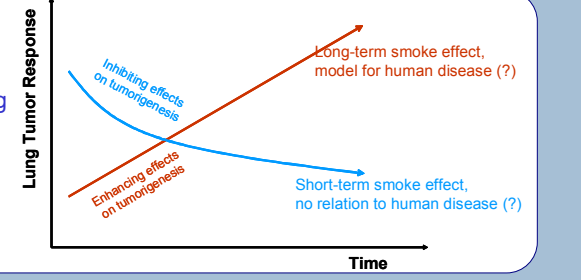
Lung Tumor Response
Time
Inhibiting effects on tumorigenesis
Enhancing effects on tumorigenesis
Long-term smoke effect, model for human disease (?)
Short-term smoke effect, no relation to human disease (?)

Materials and Methods

- Male A/J and Swiss SWR/J mice bred under specific-pathogen-free conditions (Jackson Laboratories, Maine, USA)
- Whole-body exposure
- Mainstream smoke (MS) from the Reference Cigarette 2R4F generated under ISO conditions:
 - Vapor-phase-depleted particle phase (PP) generated by passing WS through an activated charcoal filter
 - Vapor phase (VP) generated by passing WS through an electrostatic filter
- The studies were conducted in accordance with the following:
 - OECD principles of Good Laboratory Practice (1998)
 - American Association for Laboratory Animal Science Policy on the Human Care and Use of Laboratory Animals (1991)
- Endpoints (with references for methods):
 - Analytical test atmosphere characterization (Stinn et al, 2005b)
 - Body weight
 - Carboxyhemoglobin (HbCO) in blood (Stinn et al., 2005a)
 - Corticosterone in plasma (stress marker) (Stinn et al., 2005a)
 - Inflammatory cells, cytokines, and chemokines in bronchoalveolar lavage (BAL) fluid (n = 6 to 10): 5 subsequent lavage cycles with 1 ml PBS, 0.325% BSA. BAL leukocytes were differentiated by flow cytometry using antibodies against Ly6G and F4/80 (BD Biosciences) and propidium iodide counterstaining of fixed, saponin-permeabilized cells (similarly to Friedrichs et al., 2006). Markers were determined using a multiplex ELISA (Endogen SearchLight Multiplex Assay).
 - Gene expression in whole lung homogenates was performed on RNA from non-tumor tissue samples using the Agilent whole mouse genome oligo microarrays (4x44K) (pooled samples from 4 mice each).
 - K*ras* mutational analysis (Stinn et al., 2005): DNA was prepared from fresh-frozen tumor tissue or from formalin-fixed, paraffin-embedded tumor tissue using laser capture microdissection. Sequencing of the amplified PCR fragments of exons 1 and 2 was performed by Qiagen (Hilden, Germany).
 - Macroscopic counting of nodules after immersion in Tellyesniczky's fixative for 1 day followed by fixation in ethanol (Stinn et al., 2005a).
 - Microscopy of lung sections: step-serial sections per 100 (Study 1) and 300 µm (Study 2) after hematoxylin/eosin (HE) staining; lung tumor classification according to WHO guidelines (2001).

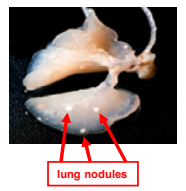
lung nodules

- Statistics:
 - Biological data: mean ± standard error (inflammatory markers: median with 25% and 75% quartiles)
 - Analytical data: mean ± standard deviation
 - In figures:
 * statistically significant at p <0.05 compared to sham
 - Lung tumor incidence: chi-square test
 - Lung tumor multiplicity: Kruskal-Wallis followed by Dunnett
 - BAL cells: one-way ANOVA followed by Tukey (BAL markers: prior transformation by ranking)
 - All other continuous data: one-way ANOVA followed by Dunnett

References

Association for the Assessment and Accreditation of Laboratory Animal Care International. 1991. American association for laboratory animal science policy on the humane care and use of laboratory animals. *Lab. Anim. Sci.* 41: 91-91.
Balansky, R., G. Ganchev, M. Iltcheva, V. E. Steele, F. D'Agostini, and S. De Flora. 2007. Potent carcinogenicity of cigarette smoke in mice exposed early in life. *Carcinogenesis* 28: 2236-2243.
Coggins, C. R. E. 2007. An Updated Review of Inhalation Studies with Cigarette Smoke in Laboratory Animals. *Internat. J. Toxicol.* 26: 331-338.
Curtin, G. M., M. A. Higuchi, P. H. Ayres, J. E. Swauger, and A. T. Mosberg. 2004. Lung tumorigenicity in A/J and rasH2 transgenic mice following mainstream tobacco smoke inhalation. *Toxicol. Sci.* 81: 26-34.
D'Agostini, F., R. M. Balansky, C. Bennicelli, R. A. Lubet, G. J. Kelloff, and De Flora S. 2001. Pilot studies evaluating the lung tumor yield in cigarette smoke-exposed mice. *Int. J. Oncol.* 18: 607-615.
De Flora, S., F. D'Agostini, R. Balansky, A. Camoirano, C. Bennicelli, M. Bagnasco, C. Cartiglia, E. Tampa, M. G. Longobardi, R. A. Lubet, and A. Izzotti. 2003. Modulation of cigarette smoke-related end-points in mutagenesis and carcinogenesis. *Mutat. Res.* 523-524: 237-252.
Finch, G. L., K. J. Nikula, S. A. Belinsky, E. B. Barr, G. D. Stoner, and J. F. Lechner. 1996. Failure of cigarette smoke to induce or promote lung cancer in the A/J mouse. *Cancer Lett.* 99: 161-167.
Friedrichs, B., E. Miert, and P. Vanscheeuwijck. 2006. Lung inflammation in rats following subchronic exposure to cigarette mainstream smoke. *Exp. Lung Res.* 32: 151-179.
Hamm, J. T., S. Yee, N. Rajendran, R. L. Morrissey, S. J. Richter, and M. Misra. 2007. Histological alterations in male A/J mice following nose-only exposure to tobacco smoke. *Inhal. Toxicol.* 19: 405-418.
Haussmann, H.-J. 2007. Smoking and Lung Cancer: Future Research Directions. *Internat. J. Toxicol.* 26: 353-364.
Hutt, J. A., B. R. Vuillemenot, E. B. Barr, M. J. Grimes, F. F. Hahn, C. H. Hobbs, T. H. March, A. P. Gigliotti, S. K. Seilkop, G. L. Finch, J. L. Mauderly, and S. A. Belinsky. 2005. Life-span inhalation exposure to mainstream cigarette smoke induces lung cancer in B6C3F1 mice through genetic and epigenetic pathways. *Carcinogenesis* 26: 1999-2009.
Mauderly, J. L., A. P. Gigliotti, E. B. Barr, W. E. Bechtold, S. A. Belinsky, F. F. Hahn, C. A. Hobbs, T. H. March, S. K. Seilkop, and G. L. Finch. 2004. Chronic inhalation exposure to mainstream cigarette smoke increases lung and nasal tumor incidence in rats. *Toxicol. Sci.* 81: 280-292.
Organization for Economic Co-operation and Development Environment Directorate. 1998. The OECD Series on Principles of Good Laboratory Practice and Compliance Monitoring - Number 1. Paris: Organization for Economic Co-operation and Development.
Schleef, R. R., P. M. Vanscheeuwijck, W. K. Schlage, J. F. Borzelleca, C. R. E. Coggins, and H.-J. Haussmann. 2006. Animal models for three major cigarette-smoke-induced diseases. In *Inhalation Toxicology (Second Edition)*, eds. H. Salem and S. A. Katz, pp. 851-873. Boca Raton: CRC Press.
Stinn, W., A. Teredesai, E. Anskeit, K. Rustemeier, G. Schepers, P. Schnell, H.-J. Haussmann, R. A. Carchman, C. R. Coggins, and W. Reininghaus. 2005. Chronic nose-only inhalation study in rats, comparing room-aged sidestream cigarette smoke and diesel engine exhaust. *Inhal. Toxicol.* 17: 549-576.
Stinn, W., A. Teredesai, P. Kuhl, C. Knorr-Wittmann, R. Kindt, C. Coggins, and H.-J. Haussmann. 2005. Mechanisms involved in A/J mouse lung tumorigenesis induced by inhalation of an environmental tobacco smoke surrogate. *Inhal. Toxicol.* 17: 263-276.
US Department of Health and Human Services. 2004. The health consequences of smoking: a report of the Surgeon General. Washington, DC: Office on Smoking and Health.
Witschi, H. 2005a. Carcinogenic activity of cigarette smoke gas phase and its modulation by beta-carotene and N-acetylcysteine. *Toxicol. Sci.* 84: 81-87.
Witschi, H. 2005b. The complexities of an apparently simple lung tumor model: The A/J mouse. *Exp. Toxicol. Pathol.* 57 Suppl 1: 171-181.
World Health Organisation. 2001. International classification of rodent tumors. The mouse. Heidelberg, Germany: Springer Verlag.

*Current address: Kessel-Lo, Belgium

American College of Toxicology, 29th Annual Meeting, Tucson, Arizona, USA, 9-12 November 2008

This study was supported in part by Philip Morris USA