Characterization of Strain A Mice as a Potential Model for Lung Emphysema

E. Van Miert[1], B. Friedrichs[2], H. Weiler[2], J. Pype[1], M. Peck[3], P. Vanscheeuwijck[1]

[1]Philip Morris Research Laboratories bvba, Leuven, Belgium, [2]Philip Morris Research Laboratories GmbH, Cologne, Germany; [3]Philip Morris International R&D, Neuchâtel, Switzerland

Introduction

Cigarette smoking is a major cause of chronic obstructive pulmonary disease (COPD). Animal models of cigarette-smoke-induced COPD, or of pathologies associated with COPD (such as lung emphysema, chronic bronchitis, and bronchiolitis), are critical to evaluate the effectiveness of new cigarette products with potentially reduced risk (Hatsukami et al., 2007; IOM 2001).

Objective

Assess the suitability of the Strain A Mouse as a model for cigarette-smoke-induced lung emphysema.

Conclusion

Overall, A/J mice exposed to mainstream cigarette smoke showed inflammatory, functional, and histological (morphometric) changes that can be associated with pulmonary emphysema. As such, the A/J mouse appears to be a useful model for cigarette-smoke-induced lung emphysema.

Materials and Methods

Animals and Cigarette Smoke Exposure

Female A/J mice (24-25 weeks old at the start of the study) exposed in whole body chambers to fresh air or to diluted mainstream smoke (MS) from the Reference Cigarette 2R4F for 2, 3, or 4 hours /day, 5 days/week at 750 µg total particulate matter (TPM)/l [corresponding to 1500, 2250, and 3000 µg TPM/l) x h] for 5 months.

Daily exposure schedule:

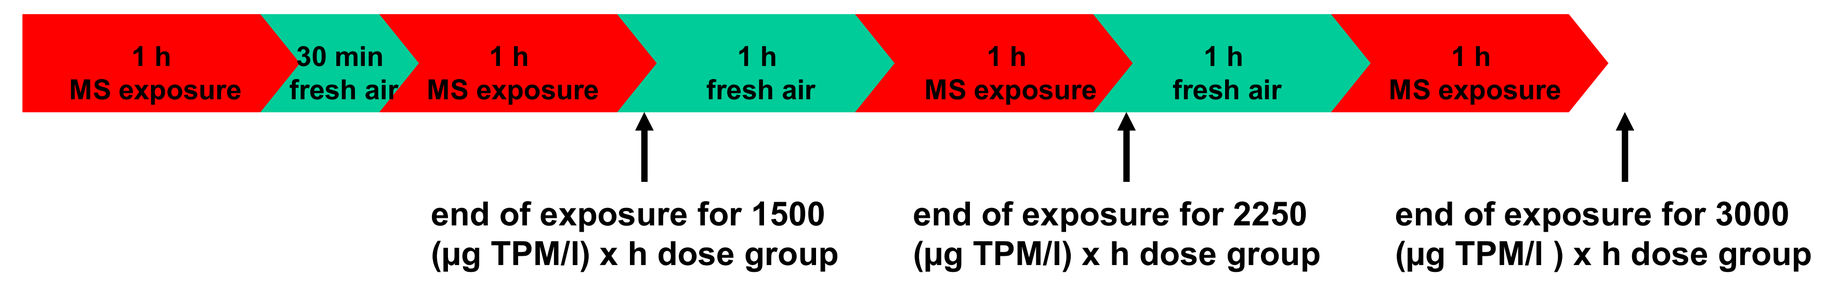
1 h MS exposure
30 min fresh air
1 h MS exposure
1 h fresh air
1 h MS exposure
1 h fresh air
1 h MS exposure
end of exposure for 1500 (µg TPM/l) x h dose group
end of exposure for 2250 (µg TPM/l) x h dose group
end of exposure for 3000 (µg TPM/l) x h dose group

Smoke Generation and Exposure Setup

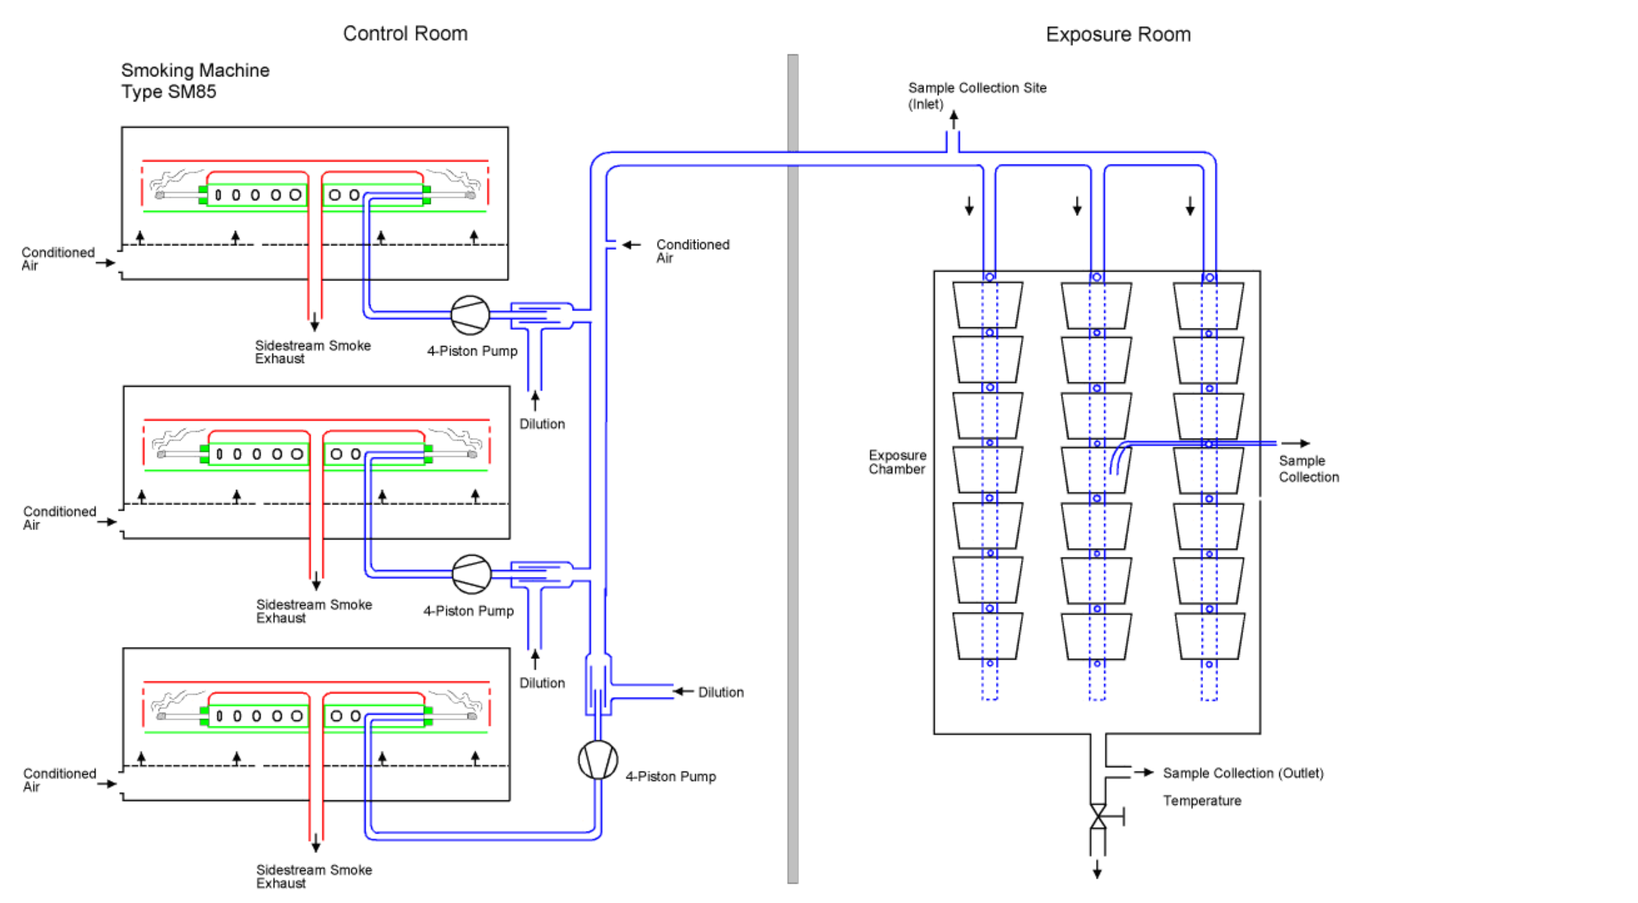
Control Room
Exposure Room
Smoking Machine Type SM85
Conditioned Air
Sidestream Smoke Exhaust
4-Piston Pump
Dilution
Exposure Chamber
Sample Collection Site (Inlet)
Sample Collection
Sample Collection (Outlet)
Temperature

Test Atmosphere Characterization and Biomonitoring

The test atmosphere was monitored throughout the study to ensure reproducible composition and correct dosing.

Parameter		Air Exposure			MS Exposure		
		Mean	SD	N	Mean	SD	N
TPM	(µg/l)	<6.7	-	95	735.0	43.0	95
carbon monoxide	(ppm)	<4.5	-	95	792.5	45.9	95
nicotine	(µg/l)	<0.11	-	18	42.57	4.44	18
formaldehyde	(µg/l)	ND			0.39	0.04	18
acetaldehyde	(µg/l)	ND			27.30	1.50	18
acrolein	(µg/l)	ND			2.03	0.10	18

Biomonitoring: Carboxyhemoglobin (HbCO) levels were measured in all groups to ensure comparable smoke uptake.

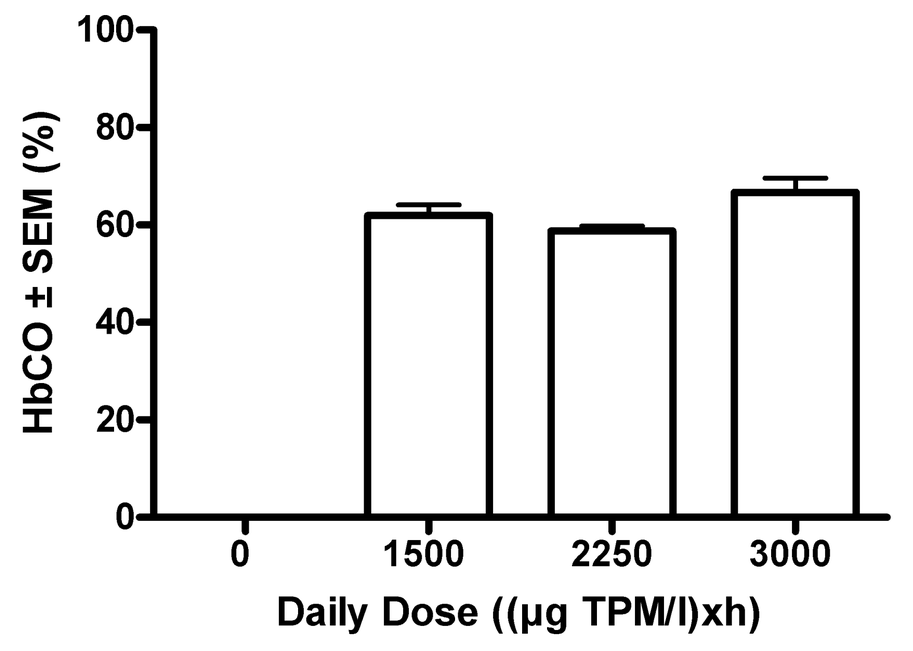
HbCO ± SEM (%)
Daily Dose ((µg TPM/l)xh)
0
1500
2250
3000

N=6

Endpoints

- The measurement of lung function and the collection of tissue samples and bronchoalveolar lavage fluid (BALF) was performed 24 h after the last exposure.
- Inflammation:
 - Flow cytometric analysis
 - FACSCanto with FACSDiva software (Becton Dickinson)
 - differentiation of free lung cells in BALF
 - measurements of activation marker expression in alveolar macrophages: CD86 (co-stimulatory molecule B7.2) and CD11b (alpha-chain of Mac-1 integrin)
 - differentiation of lymphocytes in bronchial lymph nodes (CD4- and CD8-positive T-cells, B-cells)
 - Proteins in BALF and serum
 - Rodent Multi-Analyte Profile, Rules Based Medicine, Inc., Austin, USA (www.rulesbasedmedicine.com)
 - concentration in BALF determined in the cell-free supernatant of 1st lavage cycle
- Respiratory Function:
 - Determination of respiratory mechanics: tissue elastance (H) and maximal pressure (P_{aomax})
 - Flexivent system, Scireq, Montreal, Canada (www.scireq.com) for methodological details: (http://www.pa2online.org/abstract/abstract.jsp?abid=28295)
- Morphometry – Histopathology:
 - Morphometrical evaluation of HE-stained lung slices
 - determination of mean chord length, bronchial attachments by Visiopharm®, Hørsholm, Denmark (www.visiopharm.com)
 - determination of alveolar circumference and circularity index by BIOVAYS, Marseille, France (www.biovays.com)

Statistical Analysis

One-way ANOVA followed by Dunnett post-hoc test. Data was log- or rank-transformed if required * : p<0.05

Results: Inflammation

There is clear evidence of pronounced and ongoing inflammation in the lung.

Flow Cytometric Analysis

Free Lung Cell Differentiation

Neutrophils

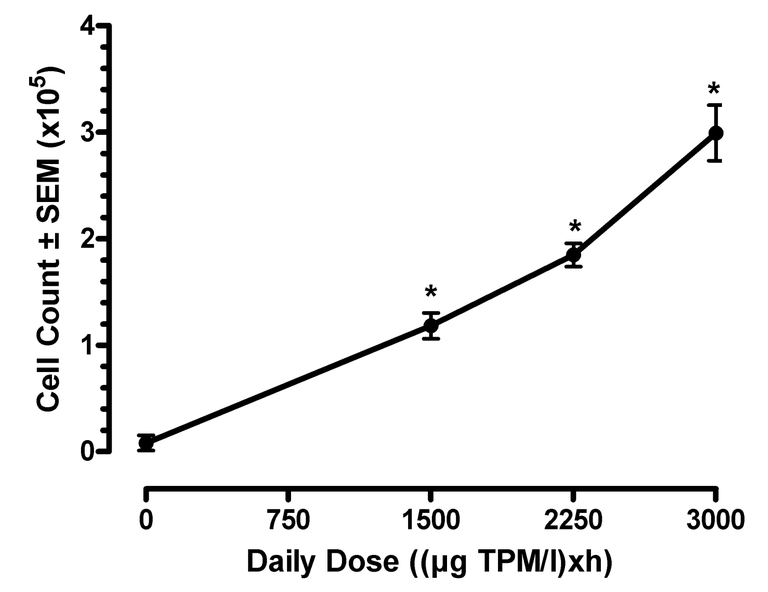
Cell Count ± SEM (x10^5)
Daily Dose ((µg TPM/l)xh)

(N =13-15)

Lymphocytes

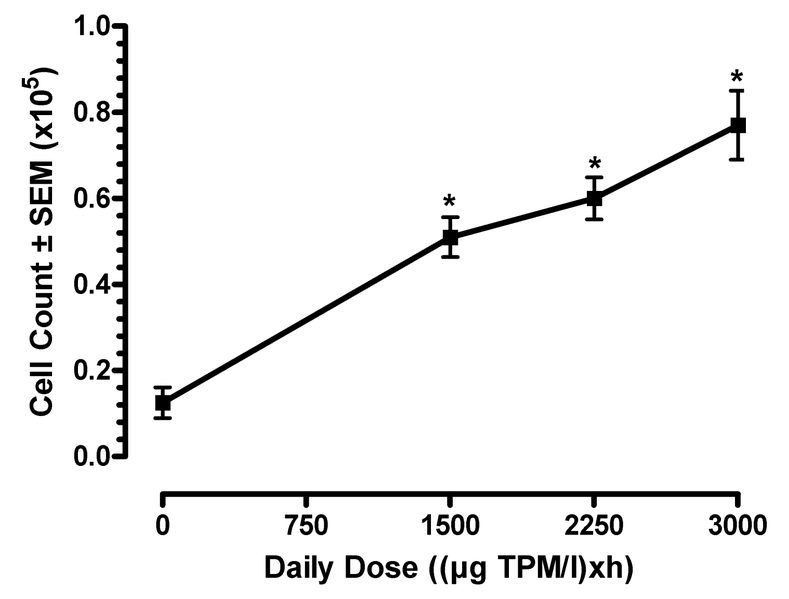
Cell Count ± SEM (x10^5)
Daily Dose ((µg TPM/l)xh)

(N =13-15)

- Marked increases in both neutrophils (37-fold) and lymphocytes (6-fold) are indicative of an ongoing inflammatory process in the lungs of MS-exposed mice

Activation Status of Alveolar Macrophages

Expression of CD11b

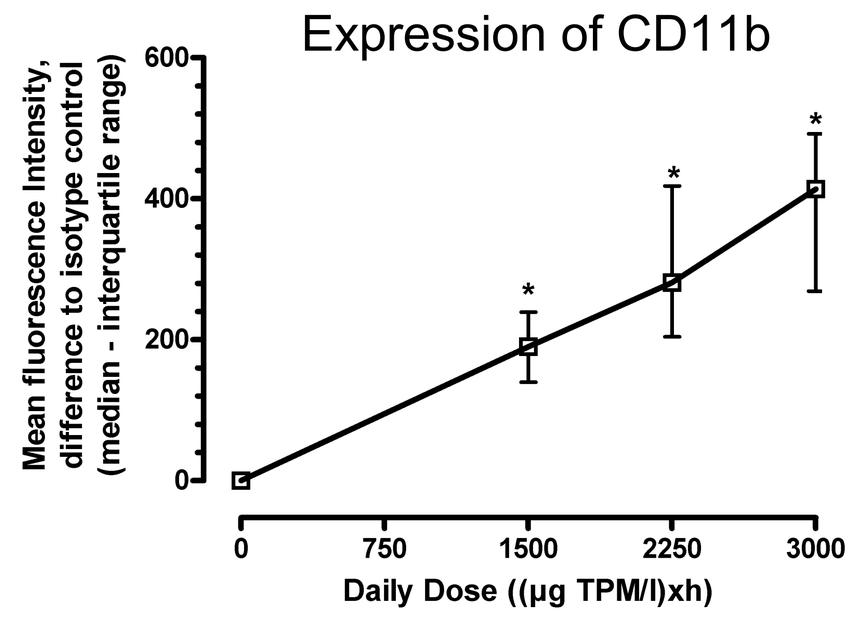
Mean fluorescence Intensity, difference to isotype control (median - interquartile range)
Daily Dose ((µg TPM/l)xh)

(N =11-13)

Expression of CD86

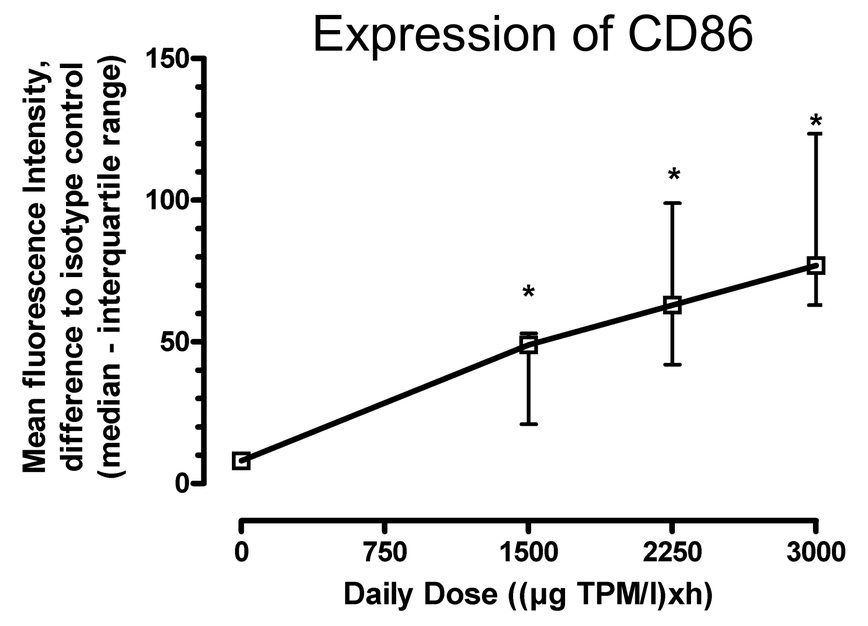
Mean fluorescence Intensity, difference to isotype control (median - interquartile range)
Daily Dose ((µg TPM/l)xh)

(N =11-13)

- Expression of CD11b (414-fold increase) and CD86 (10-fold increase) indicates marked macrophage activation in the lungs of MS-exposed mice

Lymphocyte Differentiation in Bronchial Lymph Nodes

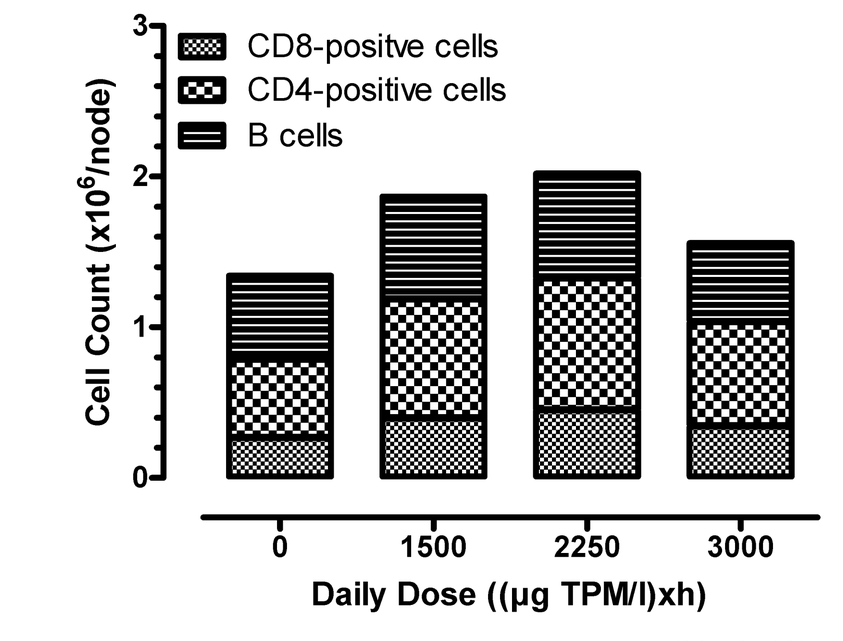
CD8-positve cells
CD4-positive cells
B cells
Cell Count (x10^6/node)
Daily Dose ((µg TPM/l)xh)

(N =9-10)

- No consistent changes in total cell numbers
- No significant changes in relative numbers of cell types
- Results suggest that the inflammation was confined mainly to the lung

Proteins in BALF and Serum

Inflammatory Mediators

Monocyte Chemoattractant Protein 1

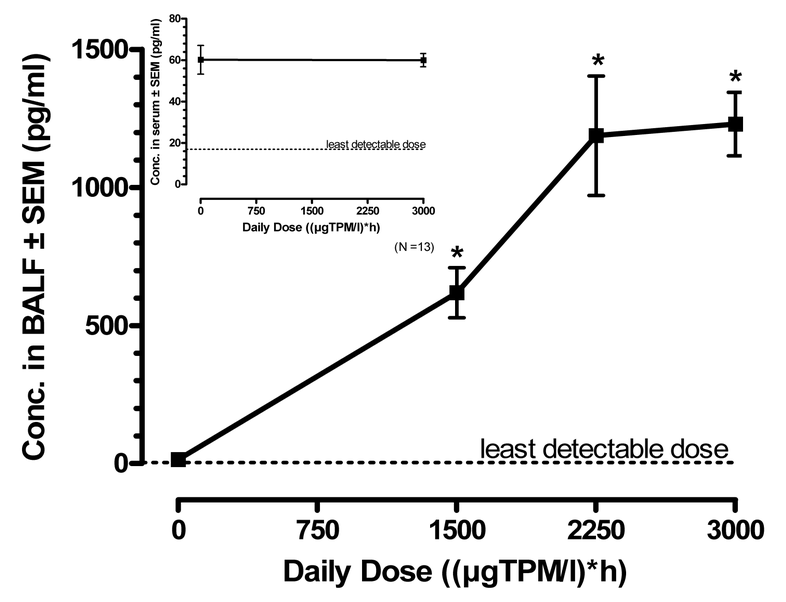
Conc. in BALF ± SEM (pg/ml)
least detectable dose
Daily Dose ((µgTPM/l)*h)

(N =13-16)

Macrophage Inflammatory Protein 2

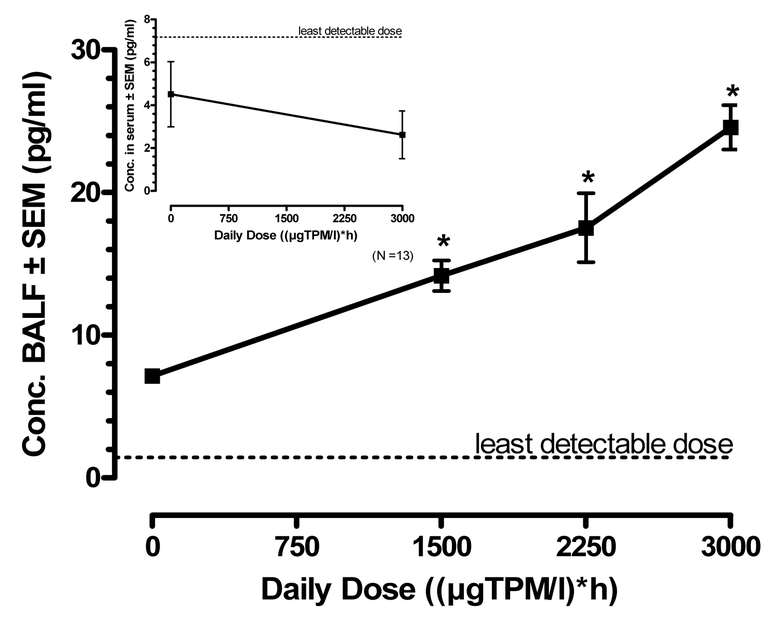
Conc. BALF ± SEM (pg/ml)
least detectable dose
Daily Dose ((µgTPM/l)*h)

(N =13-16)

- Increases in pro-inflammatory cytokines and monocyte/macrophage- and neutrophil-chemoattractant proteins in BALF, e.g., MCP1 (94-fold), MIP2 (4-fold)
- Increases observed in BALF were not paralleled by increased levels in serum, suggesting that the inflammation was confined to the lung

Other Proteins in BALF

Immunoglobulin A

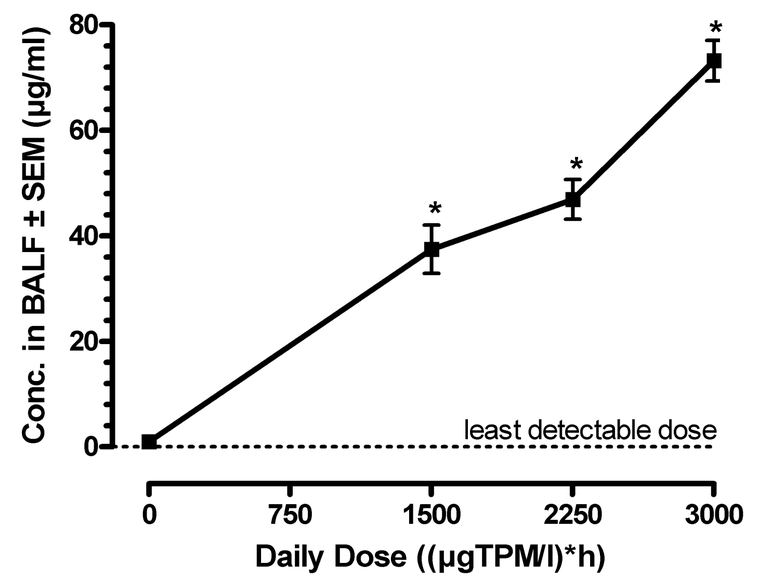
Conc. in BALF ± SEM (µg/ml)
least detectable dose
Daily Dose ((µgTPM/l)*h)

(N =13-16)

Protease (MMP9) and anti-protease (TIMP-1)

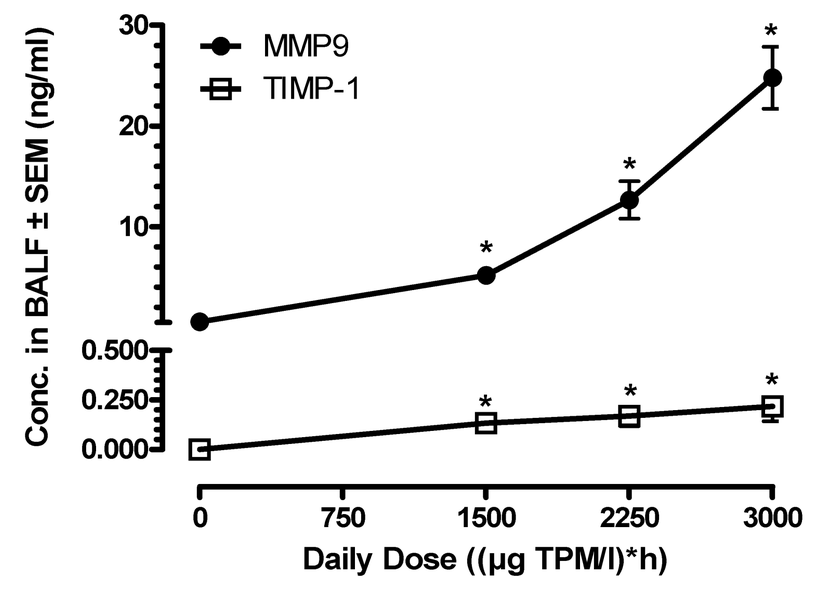
MMP9
TIMP-1
Conc. in BALF ± SEM (ng/ml)
Daily Dose ((µg TPM/l)*h)

(N =13-16)

- Pronounced increase in IgA (149-fold)—indicative of a stimulation of the humoral system
- Increases in both MMP9 and TIMP-1 (45-fold and 6-fold, respectively)—the more pronounced increase in MMP9 might be indicative of a protease-antiprotease imbalance

Results: Respiratory Function

There is clear evidence of altered lung mechanics in mice exposed to MS.

Tissue Elastance

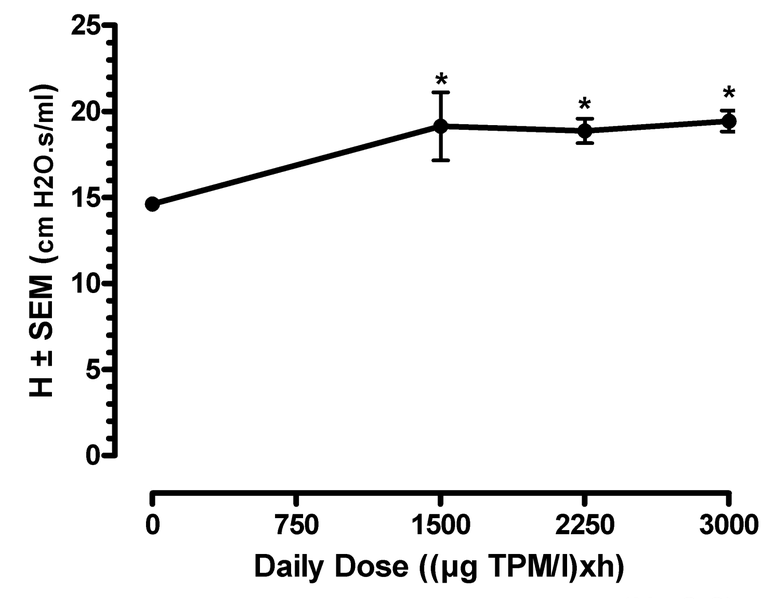
H ± SEM (cm H2O.s/ml)
Daily Dose ((µg TPM/l)xh)

(N =8-9)

- The higher tissue elastance (33% high dose vs. sham) suggests an increased rigidity of the lung in MS-exposed mice

Maximal Pressure

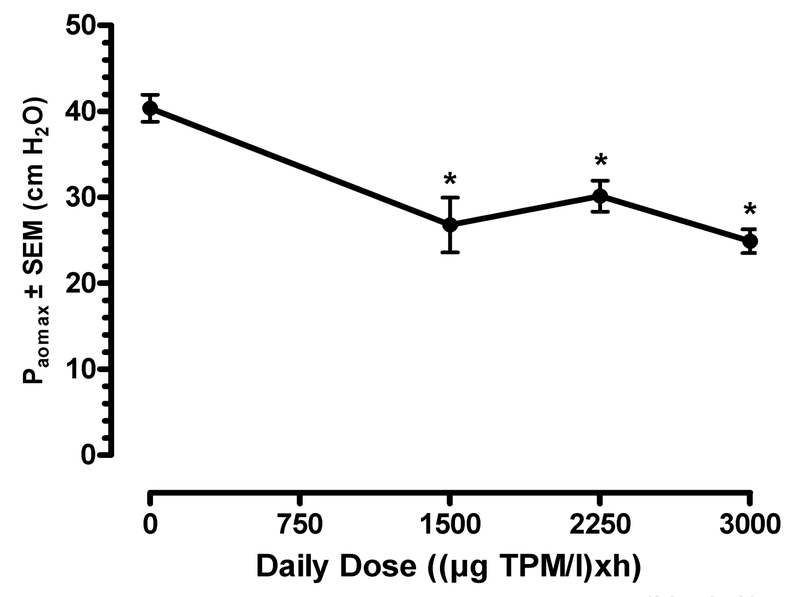
Paomax ± SEM (cm H2O)
Daily Dose ((µg TPM/l)xh)

(N =8-9)

- The lower maximal pressure (-38% high dose vs. sham) at total lung capacity indicates a loss of elastic recoil of the lung in MS-exposed mice

References

Hatsukami DK, Joseph AM, Lesage M, Jensen J, Murphy SE, Pentel PR, Kotlyar M, Borgida E, Le C, Hecht SS. Developing the science base for reducing tobacco harm. Nicotine Tob. Res. 9(4):S537-53 (2007)

Institue of Medicine. Clearing the Smoke: assessing the science base for tobacco harm reduction. Stratton, K., Shety, P., Wallace, R., and Bondurant, S. 2001. Washington, D.C., National Academy Press.

Results: Morphometry - Histopathology

There is clear evidence of altered lung morphology in mice exposed to MS.

Morphology

Exposure to Air

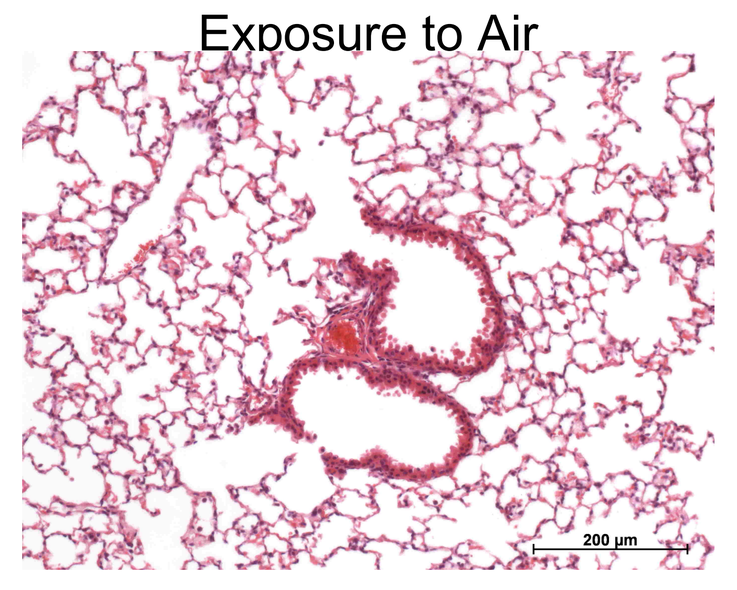
200 µm

Exposure to MS

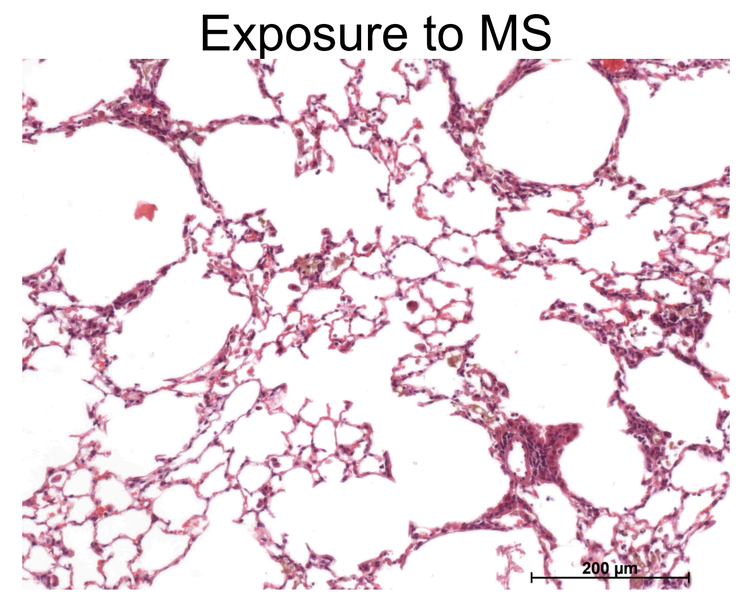
200 µm

Qualitative assessment:
- Emphysematous destruction of alveolar walls
- Peribronchiolar and perivascular infiltration by inflammatory cells
- Alveoli containing brown-pigmented alveolar macrophages

Mean Chord Length and Bronchial Attachments

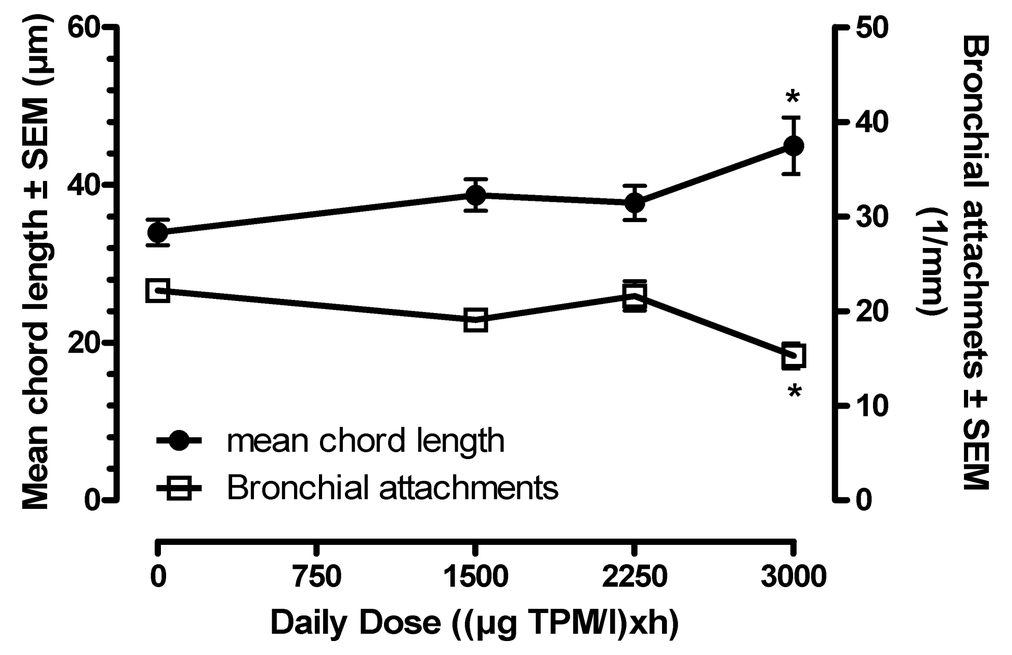
Mean chord length ± SEM (µm)
Bronchial attachmets ± SEM (1/mm)
mean chord length
Bronchial attachments
Daily Dose ((µg TPM/l)xh)

(N =10)

- In the highest MS dose group, an increased mean chord length (32% high dose vs. sham) was paralleled by a decreased number of bronchial attachments (-31% high dose vs. sham)

Alveolar Perimeter and Circularity Index

Alveolar Perimeter

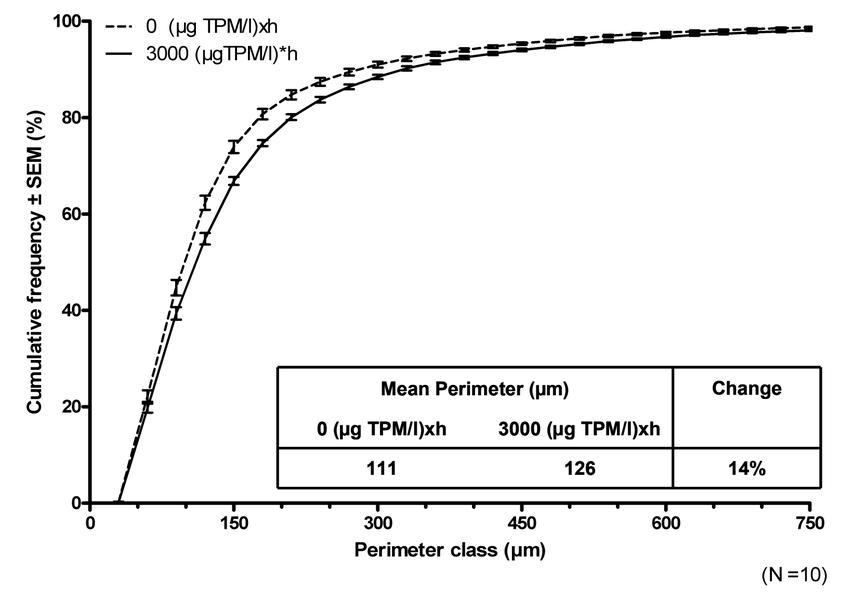
0 (µg TPM/l)xh
3000 (µgTPM/l)*h
Cumulative frequency ± SEM (%)
Mean Perimeter (µm)
Change
0 (µg TPM/l)xh
3000 (µg TPM/l)xh
111
126
14%
Perimeter class (µm)

(N =10)

Circularity Index

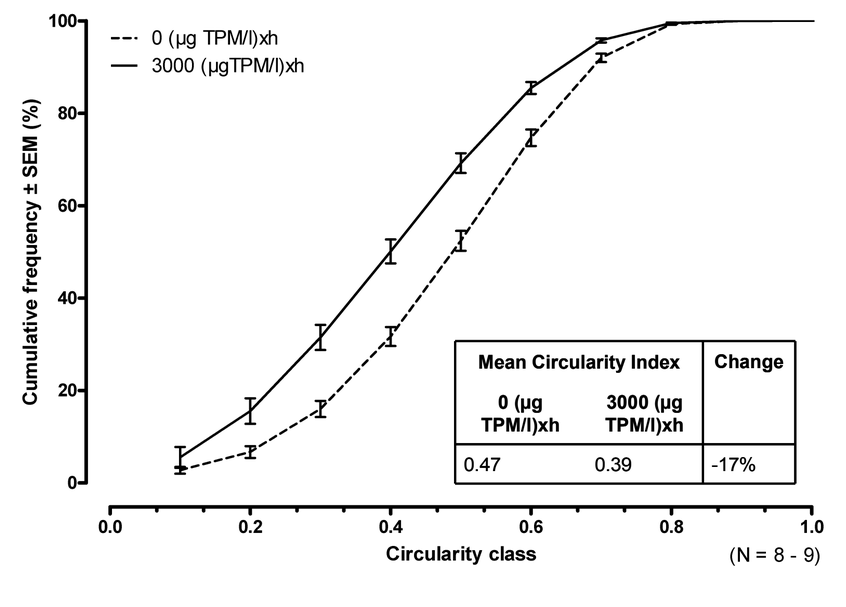
0 (µg TPM/l)xh
3000 (µgTPM/l)xh
Cumulative frequency ± SEM (%)
Mean Circularity Index
Change
0 (µg TPM/l)xh
3000 (µg TPM/l)xh
0.47
0.39
-17%
Circularity class

(N = 8 - 9)

- The distribution of the alveolar perimeters showed a clear shift towards larger perimeters in lungs of MS-exposed mice
- The distribution of the circularity indexes showed a clear shift towards lower circularity indexes, i.e., less round alveoli, in lungs of MS-exposed mice

American College of Toxicology, 29th Annual Meeting, Tucson, Arizona, USA, 9-12 November 2008

This work was supported in part by Philip Morris USA.