A One-month Study to Evaluate Biomarkers of Exposure and Effect in Smokers Switching from Conventional Cigarettes to an Electrically Heated Cigarette Smoking System

Claire Martin Leroy [1], Katarzyna Jarus-Dziedzic[2] , Jacek Ancerewicz[1], Mark Bentley[3], Dirk Lindner[1], John Magnette[1], Hugh Browne[1]
[1] Philip Morris Products S.A., Neuchâtel, Switzerland; [2] MTZ Clinical Research Ltd, Warsaw, Poland; [3] Philip Morris Research Laboratories GmbH, Köln, Germany

PMI RESEARCH & DEVELOPMENT

Introduction and Objective

Previous studies have demonstrated reductions in exposure to selected smoke constituents in smokers that switch from smoking conventional cigarettes (CC) to a third generation Electrically Heated Cigarette Smoking System (EHCSS) Series K under controlled smoking conditions [1]. This smoking device is used in combination with specially designed EHCSS-K6 cigarettes.

The aim of this study was to investigate changes in a panel of biomarkers of exposure (BoExp) and of candidate biomarkers of effect (BoEff) in subjects that switch from smoking conventional cigarettes to EHCSS-K6 cigarettes for one month under real-life conditions.

Materials and Methods

This clinical investigation was a single-centre, controlled, randomised, open-label, two-arm study. Caucasian smokers aged from 30 to 60 years were enrolled and randomised to one of two groups: those who were to continue to smoke conventional cigarettes (CC) and those who were to switch to smoking the EHCSS-K6 cigarette for one month. A randomisation ratio of 3:1 (EHCSS-K6:CC) was used. Blood and urine samples were taken from subjects and selected BoExp and BoEff were compared between study groups at the end of the study.

This study was conducted in Poland from October 2007 to April 2008 in accordance with the principles of International Committee on Harmonisation (ICH) Good Clinical Practice [2].

Results - Demographics

A total of 338 subjects were enrolled, of which 316 were randomised to either the CC study arm (N = 79) or to the EHCSS-K6 study arm (N = 237) as presented in the figure below. The demographic data was comparable in both study arms with approximately 50% male and 50% female subjects with a mean age of 44 years (range 30–59). Subjects in the CC study arm had the same median daily cigarette consumption at baseline and at the end of study (24 cigarettes per day), in contrast to subjects in the EHCSS-K6 arm who increased from a median of 23 to 38 cigarettes over the course of the study.

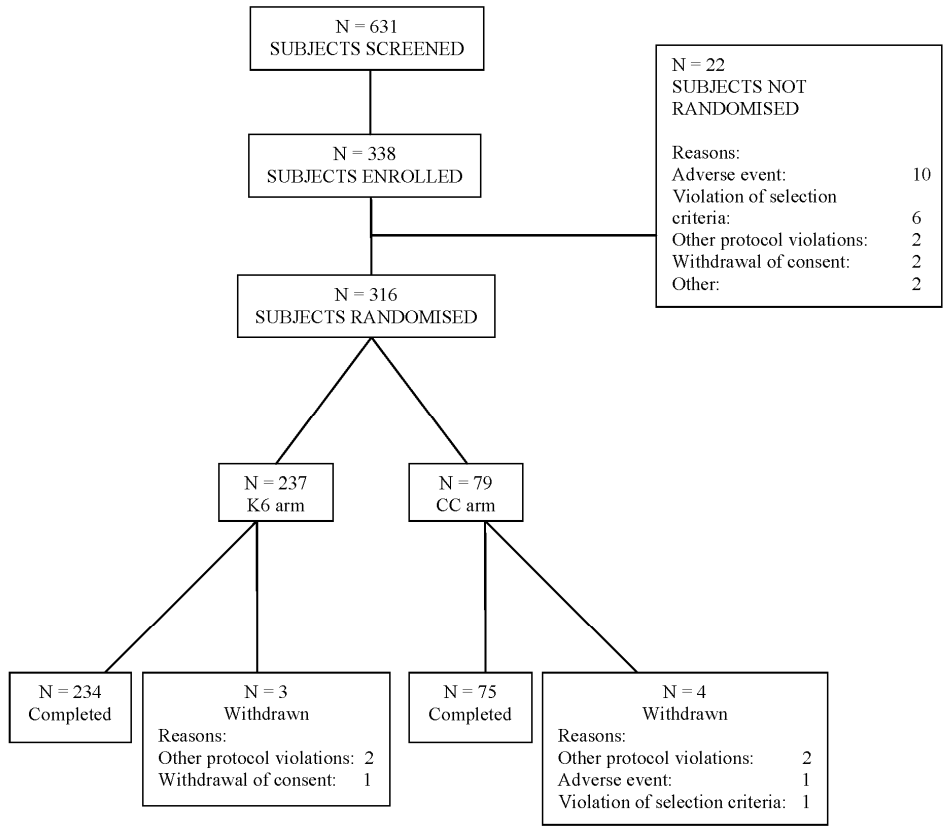
N = 631
SUBJECTS SCREENED
N = 338
SUBJECTS ENROLLED
N = 22
SUBJECTS NOT RANDOMISED
Reasons:
Adverse event: 10
Violation of selection criteria: 6
Other protocol violations: 2
Withdrawal of consent: 2
Other: 2
N = 316
SUBJECTS RANDOMISED
N = 237
K6 arm
N = 79
CC arm
N = 234
Completed
N = 3
Withdrawn
Reasons:
Other protocol violations: 2
Withdrawal of consent: 1
N = 75
Completed
N = 4
Withdrawn
Reasons:
Other protocol violations: 2
Adverse event: 1
Violation of selection criteria: 1

Results – Biomarkers of Effect

For four BoEff, statistically significant differences ($p<0.05$) were found between the study arms at the end of the study, but for the remaining BoEff no such differences were detected. An overview of this is provided in Table 1. The changes seen for the smokers of EHCSS-K6 were generally in a direction consistent with published data on smoking cessation [3]. It should be noted that all biomarkers were measured in plasma/serum except where it is specified that they were measured in urine.

Table 1: Overview of Biomarker Data

Statistically significant (p<0.05) difference at study end	Statistically significant (p<0.05) changes from baseline in study arms	No indication of change in EHCSS-K6 or CC study arms
HDL Cholesterol ↑↑ Red blood cell count ↓↓ Haemoglobin ↓↓ Haematocrit ↓↓	HDL Cholesterol – EHCSS-K6 ↑ Red blood cell count – EHCSS-K6 ↓ Haemoglobin –EHCSS-K6 ↓ Haematocrit –EHCSS-K6 ↓ LDL cholesterol – EHCSS-K6 ↓ 11-dehydrothromboxane (urine) – EHCSS-K6 ↓ hs- C-Reactive Protein – EHCSS-K6 ↓* White blood cell count – EHCSS-K6 ↓ Von Willebrand factor –EHCSS-K6 ↓ CC ↓ Oxidised LDL cholesterol – EHCSS-K6 ↓ CC ↓ ADP-induced platelet aggregation – EHCSS-K6 ↑ CC ↑	Total cholesterol Fibrinogen Homocysteine Interleukin-6 8-epi-prostaglandin - F2α (urine) Myeloperoxidase #

* Data from hs-C-reactive protein should be interpreted with caution as there was very high variability observed for this biomarker;
The mean change from baseline for myeloperoxidase was found to be different in the EHCSS-K6 study arm from that in the CC study arm;
↑↑ higher in EHCSS-K6 arm than in CC arm at end of study; ↑ increased from baseline to end of study in EHCSS-K6 arm; ↓↓ lower in EHCSS-K6 arm than in CC arm at end of study; ↓ decreased from baseline to end of study in K6 arm

Descriptive statistics for the four biomarkers for which differences were found between study arms at study end (HDL-cholesterol, red blood cell count (RBC), haemoglobin and haematocrit) are presented in Table 2.

Table 2: Summary of Selected BoEff Data

		EHCSS-K6		CC	
Parameter	Time point	N	Mean (SD)	N	Mean (SD)
HDL Cholesterol (mg/dl)	Baseline	236	59.0 (16.3)	77	61.5 (16.3)
	End of study	235	63.9 (17.3) *	75	62.3 (16.1) NS
	Change	235	5.0 (8.5)	75	0.9 (7.1)
RBC count (T/l)	Baseline	236	4.56 (0.41)	77	4.55 (0.39)
	End of study	235	4.48 (0.41) *	75	4.54 (0.39) NS
	Change	235	-0.083 (0.175)	75	-0.016 (0.143)
Haemoglobin (g/dl)	Baseline	236	14.29 (1.16)	77	14.28 (1.28)
	End of study	235	14.00 (1.18) *	75	14.21 (1.32) NS
	Change	235	-0.29 (0.51)	75	-0.06 (0.42)
Haematocrit (%)	Baseline	236	42.67 (3.16)	77	42.58 (3.41)
	End of study	235	41.75 (3.08) *	75	42.44 (3.50) NS
	Change	235	-0.92 (1.46)	75	-0.10 (1.26)

*: $p<0.05$ EHCSS-K6 end of study value is statistically significantly different from CC end of study value and different from baseline ; NS: not statistically significantly different from baseline

Results - Biomarkers of Exposure

Six BoExp for smoke constituents were measured in this study. These were carboxyhaemoglobin, total NNAL (4-(methylnitrosamino)-1-(3-pyridyl)-1-butanol), nicotine equivalents, total 1-hydroxypyrene, 2-aminonaphthalene and 4-aminobiphenyl. For these BoExp, at study end, levels were significantly lower ($p<0.05$) in the EHCSS-K6 arm than in the CC arm. Carboxyhaemoglobin and total NNAL data are presented below.

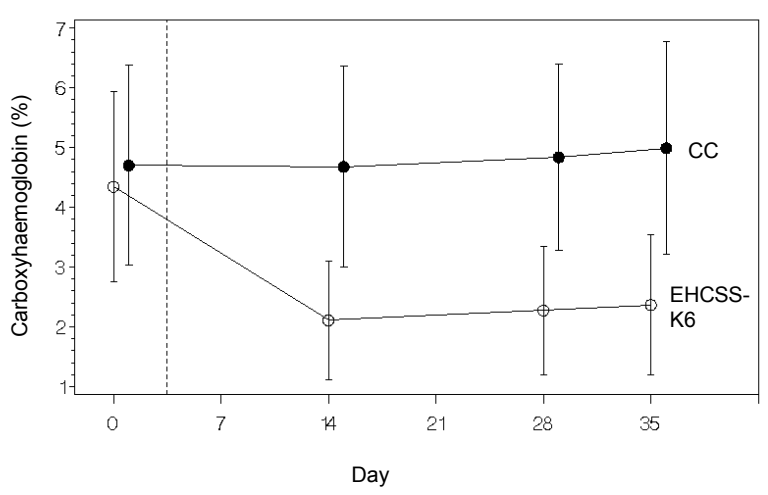
Carboxyhaemoglobin (%)
Carboxyhaemoglobin (%)
1
2
3
4
5
6
7
0
7
14
21
28
35
Day
CC
EHCSS-K6

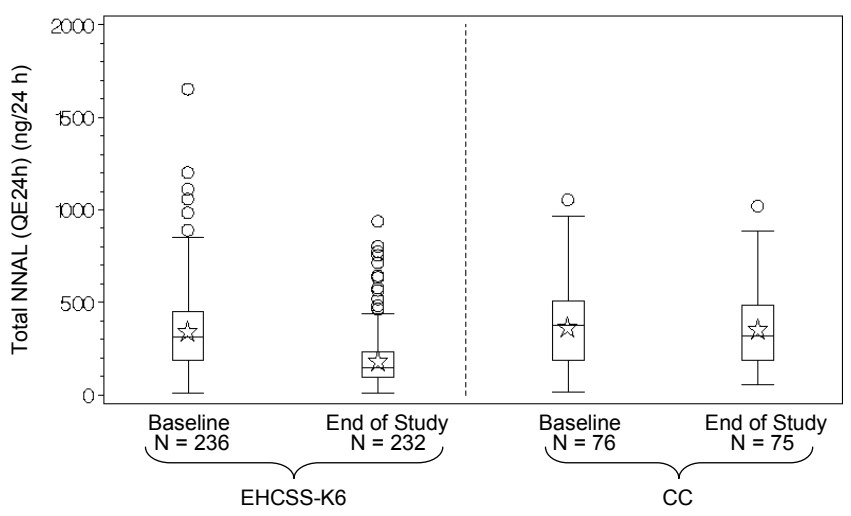
Urinary Excretion of Total NNAL (ng/24h)
Total NNAL (QE24h) (ng/24 h)
0
500
1000
1500
2000
Baseline N = 236
End of Study N = 232
Baseline N = 76
End of Study N = 75
EHCSS-K6
CC

Summary and Conclusions

This study indicates that it is feasible to detect significant changes in all BoExp and in some BoEff after smokers switch from conventional cigarettes to EHCSS-K6 cigarettes for a one-month period. This is despite an increase in daily cigarette consumption over the course of the study.

For biomarkers for which no differences were detected, there are several possible explanations such as insufficient study duration, higher than predicted variability, or lack of product effect on biomarker.

Inclusion of BoExp and BoEff in clinical studies is therefore a valuable component of an integrated product risk assessment framework for potentially reduced-risk tobacco products.

References

1 Frost-Pineda K, Zedler BK, Oliveri D, Feng S, Liang Q, Roethig HJ. Short-term clinical exposure evaluation of a third-generation electrically heated cigarette smoking system (EHCSS) in adult smokers. Regul Toxicol Pharmacol. 2008 Nov;52(2):118-21.
2 International Conference on Harmonisation Guideline for Good Clinical Practice E6. Step 4, June 1996 (www.ich.org).
3 Allen S, Hatsukami D, Gorsline J. Cholesterol changes in smoking cessation using the transdermal nicotine system. Prev Med. 1994 Mar;23(2):190-6.

Society for Research on Nicotine and Tobacco 13th Annual Meeting, Dublin, Ireland, 27-30 April 2009. Correspondence: claire.martinleroy@pmintl.com