Cigarette smoke enhances abdominal aortic aneurysm formation in angiotensin II-treated Apolipoprotein E-deficient mice

Stolle K.[1], Berges A.[2], Lietz M.[1], Lebrun S.[3], Schleef R.[4], Wallerath T.[1];
[1] Philip Morris Research Laboratories GmbH, Cologne, Germany ; [2] Philip Morris Research Laboratories bvba, Leuven, Belgium;
[3] Philip Morris Products S. A., PMI Research & Development, Neuchatel, Switzerland; [4] Altria Client Services, Richmond, VA, USA

PMI RESEARCH & DEVELOPMENT

Introduction and Objective

Apolipoprotein E-deficient (ApoE-/-) mice develop a pronounced hyperlipidemia, because the liver is not able to recognize and clear cholesterol and lipoprotein from blood. These mice consequently develop fibrous arterial plaques (corresponding to human atherosclerotic alterations and their distribution), particularly at the root, inner curvature, and branches of the aorta[(a,b)].

In previous studies[(c,d)], we showed that ApoE-/- mice develop atherosclerosis in response to cigarette mainstream smoke (MS), a risk factor for atherosclerosis in humans.

Angiotensin II (A-II) has been shown to induce abdominal aortic aneurysm (AAA) formation in mouse models. It is the major vasoconstrictive effector peptide produced by the renin angiotensin system, which causes oxidative stress and leads to hypertension. A-II has been shown to accelerate the pathogenesis of atherosclerosis, vascular remodeling, and hypertensive cardiomyopathy in both humans and animals[(e,f,g)].

In this study, we investigated the following:

- ❑ Influence of MS exposure on A-II-treated ApoE-/- mice on the abdominal aorta by analyzing AAA occurrence.
- ❑ Underlying mechanisms of AAA formation by analyzing several matrix metalloproteinases (MMPs) and their inhibitors (tissue inhibitors of matrix metalloproteinases [TIMPs]).

Materials and Methods

The in vivo study was performed at PHILIP MORRIS Research Laboratories bvba, Leuven, Belgium. Care and use of the animals was in conformity with 'The Guide for the Use and Care of Laboratory Animals' published by the US National Institute of Health[(h)]. All animal experiments were approved by the Institutional Animal Care and Use Committee (IACUC).

Male ApoE-/- mice (8-10 weeks old, Taconic) were fed a normal chow diet (Harlan Teklad) and implanted with osmotic mini-pumps (Alzet 2004) containing A-II or NaCl. Pumps were replaced after three weeks and the mice were exposed to MS from the Reference Cigarette 2R4F[(i)] or to fresh air (sham) for four weeks.

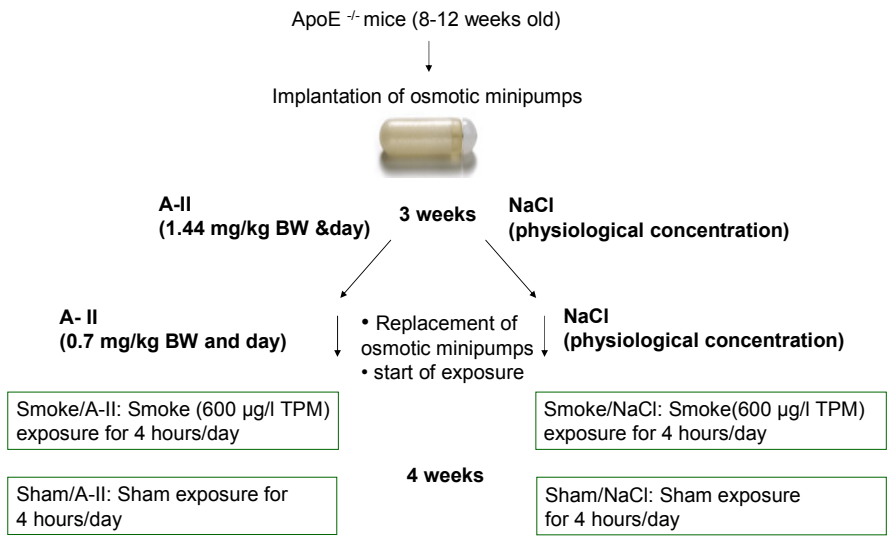
ApoE -/- mice (8-12 weeks old)
Implantation of osmotic minipumps
A-II
(1.44 mg/kg BW &day)
3 weeks
NaCl
(physiological concentration)
A- II
(0.7 mg/kg BW and day)
• Replacement of osmotic minipumps
• start of exposure
NaCl
(physiological concentration)
Smoke/A-II: Smoke (600 µg/l TPM) exposure for 4 hours/day
Smoke/NaCl: Smoke(600 µg/l TPM) exposure for 4 hours/day
4 weeks
Sham/A-II: Sham exposure for 4 hours/day
Sham/NaCl: Sham exposure for 4 hours/day

At the end of the 30-day inhalation period, animals were sacrificed and the abdominal aortas were photographed. Minimum and maximum diameter of each vessel was determined. If dilatation of the vessel wall exceeded 150% of the normal diameter, the vessel was considered to have an AAA. Vessels were then cut into different parts for extraction of RNA and protein.

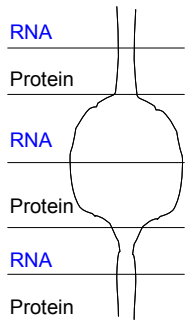
RNA
Protein
RNA
Protein
RNA
Protein

RNA was extracted with RNA lysis buffer (Quiagen) after treatment with RNAlater® and analyzed for differential gene expression (qRT-PCR) of MMPs and TIMPs.
Protein was extracted with CHAPS-buffer and analyzed for MMP activity with gelatinase zymograms (Biorad).

Results: Occurrence of Aneurysms

AAA Examples

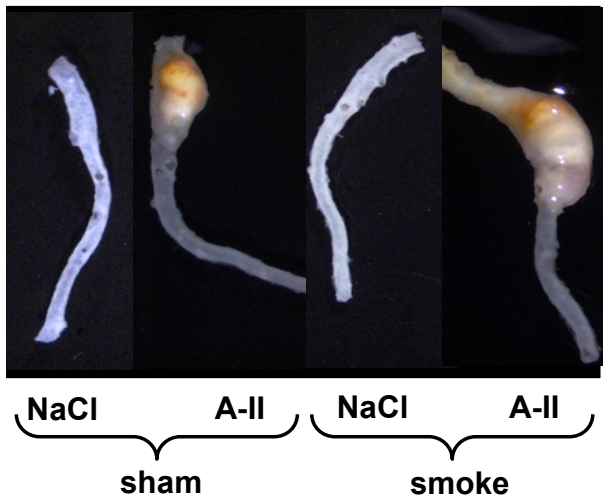
NaCl
A-II
NaCl
A-II
sham
smoke

AAA Incidence

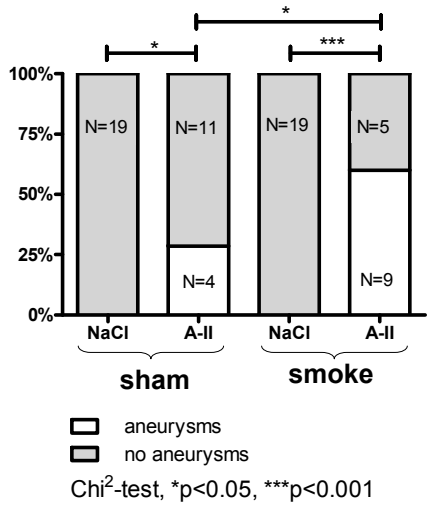
*
*

100%
75%
50%
25%
0%
N=19
N=11
N=4
N=19
N=5
N=9
NaCl
A-II
NaCl
A-II
sham
smoke
aneurysms
no aneurysms

Chi^2-test, *p<0.05, ***p<0.001

- ❑ NaCl-treated mice: no AAAs, irrespective of MS exposure.
- ❑ A-II-treated mice: AAAs in 26.7% of sham-exposed animals and in 64.3% of MS-exposed animals.

There were 37.6% more AAAs seen in MS-exposed mice, suggesting that MS potentiates the effect of A-II on AAA formation in this model. This is in line with epidemiologic evidence[(j)], which shows a strong association of smoking with risk of AAA.

Acknowledgement

This work was supported in part by Philip Morris USA, Inc. prior to the spin-off of Philip Morris International, Inc. by Altria Group, Inc. on March 28, 2008.

Results: Expression of MMPs and TIMPs

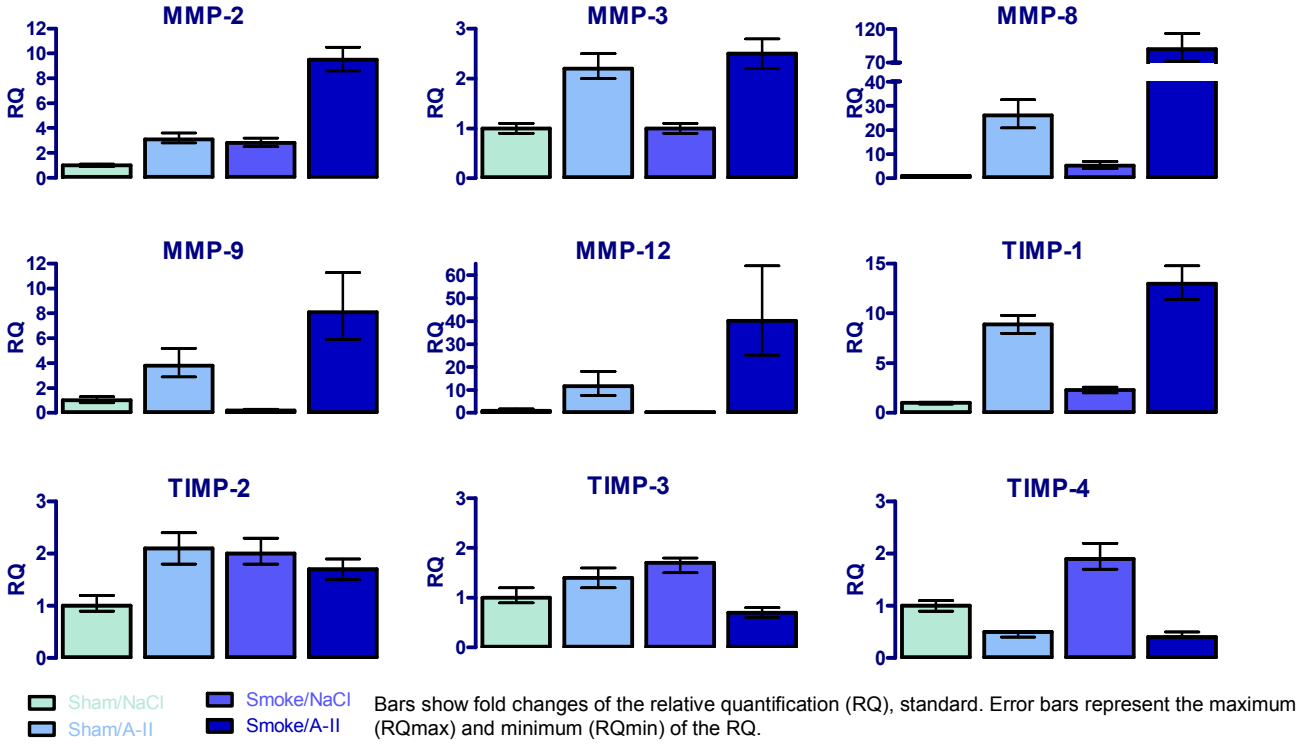
MMP-2
MMP-3
MMP-8
MMP-9
MMP-12
TIMP-1
TIMP-2
TIMP-3
TIMP-4
RQ
Sham/NaCl
Sham/A-II
Smoke/NaCl
Smoke/A-II

Bars show fold changes of the relative quantification (RQ), standard. Error bars represent the maximum (RQmax) and minimum (RQmin) of the RQ.

- ❑ Highest up-regulation of MMPs in A-II treated mice exposed to MS.
- ❑ Smoke effect: MMP-2, MMP-8, and TIMP-1.

Gene expression data correlate with the occurrence of AAA in this model and further support a mechanism for the potentiation of an A-II effect on AAA formation by MS.

Gene	Smoke Effect	A-II Effect	Interaction of Smoke and A-II
MMP-2	***	***	
MMP-3		***	
MMP-8	**	***	
MMP-9		***	
MMP-12		***	
TIMP-1	*	***	
TIMP-2			* $
TIMP-3			** $
TIMP-4		***	

Data analyzed with Two-way ANOVA for MS and A-II effect followed by one-way ANOVA when a significant interaction was detected.

Red: up-regulated

Green: down-regulated.

$ A significant interaction was observed, but no significances in the one-way ANOVA occurred.

Results: Activity of MMP-2 and MMP-9

Zymography Example

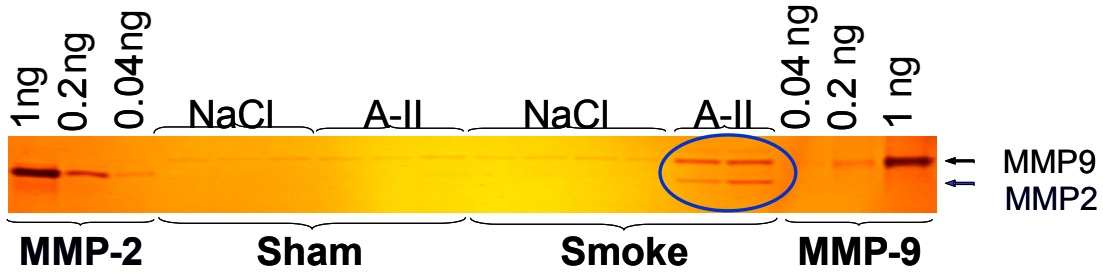
1ng
0.2ng
0.04ng
NaCl
A-II
NaCl
A-II
0.04 ng
0.2 ng
1 ng
MMP9
MMP2
MMP-2
Sham
Smoke
MMP-9

Semiquantitative Zymograpic Analysis

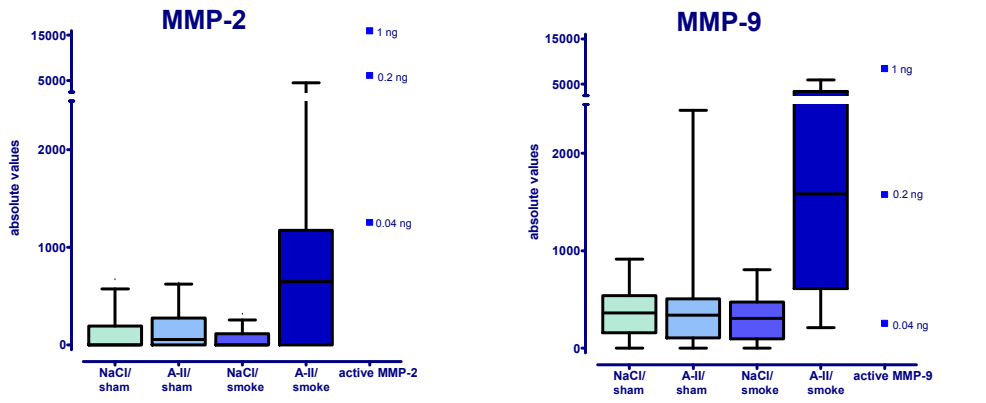
MMP-2
MMP-9
15000
5000
2000
1000
0
absolute values
1 ng
0.2 ng
0.04 ng
NaCl/ sham
A-II/ sham
NaCl/ smoke
A-II/ smoke
active MMP-2
active MMP-9
Sham/NaCl
Sham/A-II
Smoke/NaCl
Smoke/A-II

Bars represent the median with 1st and 3rd quartiles; error bars the minimum and maximum. Statistical evaluation was performed by non-parametric two-way ANOVA, followed by one-way ANOVA. For MMP-2 and MMP-9 a significant interaction (p<0.01) was observed. The A-II/smoke group showed higher activity compared to A-II/sham (p<0.05 for MMP-2, p<0.001 for MMP-9) as well as compared to NaCl/smoke (p<0.01 for MMP-2, p<0.001 for MMP-9).

- ❑ Up-regulation only in A-II-treated mice exposed to MS.

These results suggest a role for MMP-2 and MMP-9 in aneurysm formation, as also described for the human situation[(k,l)].

Conclusion

Cigarette smoke enhances abdominal aortic aneurysm formation in angiotensin II-treated Apolipoprotein E-deficient mice.

- ❑ This new animal model is suitable for studying mechanisms of cigarette-smoke-induced aortic aneurysms.
- ❑ Results suggest that expression and activity of matrix metalloproteinases may be part of the underlying mechanism of abdominal aortic aneurysm formation in this model.

References

[a] Breslow: *Science*. 1996 May 3;272(5262):685-8
[b] Nakashima *et al*.: *Arterioscler Thromb*. 1994 Jan;14(1):133-40
[c] Schroeter *et al*.: *J* J Vasc Res. 2008;45(6):480-92
[d] von Holt *et al*.: *Atherosclerosis,* in press
[e] Daugherty *et al*.: Trends Cardiovasc Med. 2004 Apr;14(3):117-20
[f] Kon *et al*.: *Curr Opin Nephrol Hypertens*. 2004 May;13(3):291-7
[g] Brasier *et al*.: *Arterioscler Thromb Vasc Biol*. 2002 Aug 1;22(8):1257-66
[h] NIH publication, NIH 85-23, revised 1996
[i] University of Kentucky, Kentucky Tobacco Research and Development Center:The Reference Cigarette, Lexington: The University of Kentucky Printing Services, 2003
[j] The 2004 United States Surgeon General's Report: The Health Consequences of Smoking. N.S.W.Public Health Bull. 2004; 15: 107,
[k] Goodall S *et al*.: *Circulation*. 2001 Jul 17;104(3):304-9
[l] Thompson *et al*.: *J.Clin.Invest.* 1995; 96: 318-326

Keystone Symposia: Dissecting the Vasculature: Function, Molecular Mechanisms and Malfunction (J8), Feb 24 - Mar 1, 2009, Vancouver, British Columbia