Cigarette smoke increases the adhesion of human monocytes to endothelial cells

Lietz M[1], Berger C[1], Lammerich H-P[1], Steffen Y[1], Stolle K[1], Lebrun S[2], Schueller J. [1], and Wallerath T[1]
[1]Philip Morris Research Laboratories GmbH, Cologne, Germany
[2]Philip Morris Products SA, Neuchâtel, Switzerland

Objective

Cigarette smoking is a well-known risk factor for the development and progression of atherosclerosis. However, the mechanisms involved are not well understood. One early step in the initiation of atherosclerosis is the adhesion of monocytes to the vascular endothelium[a)]. It is known from the literature that cigarette smoke (CS) stimulates monocytes directly in the bloodstream[b)] and induces the expression of adhesion molecules on the surface of endothelial cells[c)].

The objective of this study was to establish an *in vitro* assay which induces increased expression of adhesion molecules in human umbilical vein endothelial cells (HUVECs) after treatment with CS resulting in an increased adhesion rate of human monocytic cells (Mono Mac 6 [MM6])[d)] to HUVECs.

End points:
1) Expression of adhesion molecules in HUVECs after exposure to CS
2) Binding untreated MM6 cells to CS-exposed HUVECs and the impact of blocking antibodies.
3) Binding of CS-exposed MM6 cells to untreated HUVECs
4) Up-regulation of genes in CS-exposed HUVECS

Methods

CS generation
Mainstream CS was generated from the Reference Cigarette 2R4F (University of Kentucky, 2003)[e)] on a Borgwaldt RM 20-port smoking machine. CS was then bubbled through 36 ml phosphate-buffered saline (PBS) at room temperature to obtain a CS stock solution (smoke-bubbled PBS [sbPBS]). The sbPBS was diluted with cell culture medium to obtain concentrations of 0.045 and 0.09 puffs/ml. Time from preparation of stock solution to treatment of cells was $\leq$15 min.

Direct treatment of HUVECs and MM6 cells
Cells were treated with sbPBS (0.045 or 0.09 puffs/ml) or with PBS alone as control (Co) for 2 or 4h.

Indirect treatment of HUVECs
MM6 cells were treated with sbPBS (0.045 or 0.09 puffs/ml) for 2h then centrifuged (800 g, 7 min); the resulting supernatant was frozen at –80°C. On the day of experiment, the MM6 supernatant was thawed ($\leq$15 min, 37°C) and transferred to HUVECs for 2 or 4h.

Gene expression analysis
HUVECs were harvested and total RNA was prepared. Samples were analyzed in-house by qRT-PCR or sent to Microarray Facility (Tübingen, Germany) for gene array analysis (Affymetrix Human Genome U133 2.0).

Adhesion assay
1) HUVECs were cultured in growth medium for 3 days till confluency.
2)* HUVECs were incubated with MM6 supernatant (0.09 puffs/ml for 4h; see indirect treatment)
- Additional step with blocking antibodies: HUVECs were blocked for 1h with 1% BSA containing PBS and incubated with antibodies (10 µg/ml) for 30 min then washed with PBS (3x).
 - vascular adhesion molecule-1 (VCAM-1)
 - intracellular adhesion molecule-1 (ICAM-1)
 - endothelial selectin (E-Selectin)

3) MM6 cells were labeled with CellTracker Orange CMTMR (Molecular Probes, cat. no. C2927) before adding to HUVECs (45 min, 37°C).
4) Nucleus staining with Hoechst-Dye (Fluka, cat. no. 14533) was performed for all cells.
5) Bound rate of MM6 cells was determined with an ArrayScan VTI HCS Reader (Cellomics, Pittsburgh, PA).

*Step 2 with CS-treated MM6 cells: MM6 cells were treated directly with sbPBS (0.045 and 0.09 puffs/ml) for 2h.

Results

1) Expression of adhesion molecules in HUVECs after exposure to CS (direct and indirect treatment)—qtPCR

Direct treatment | **Indirect treatment**

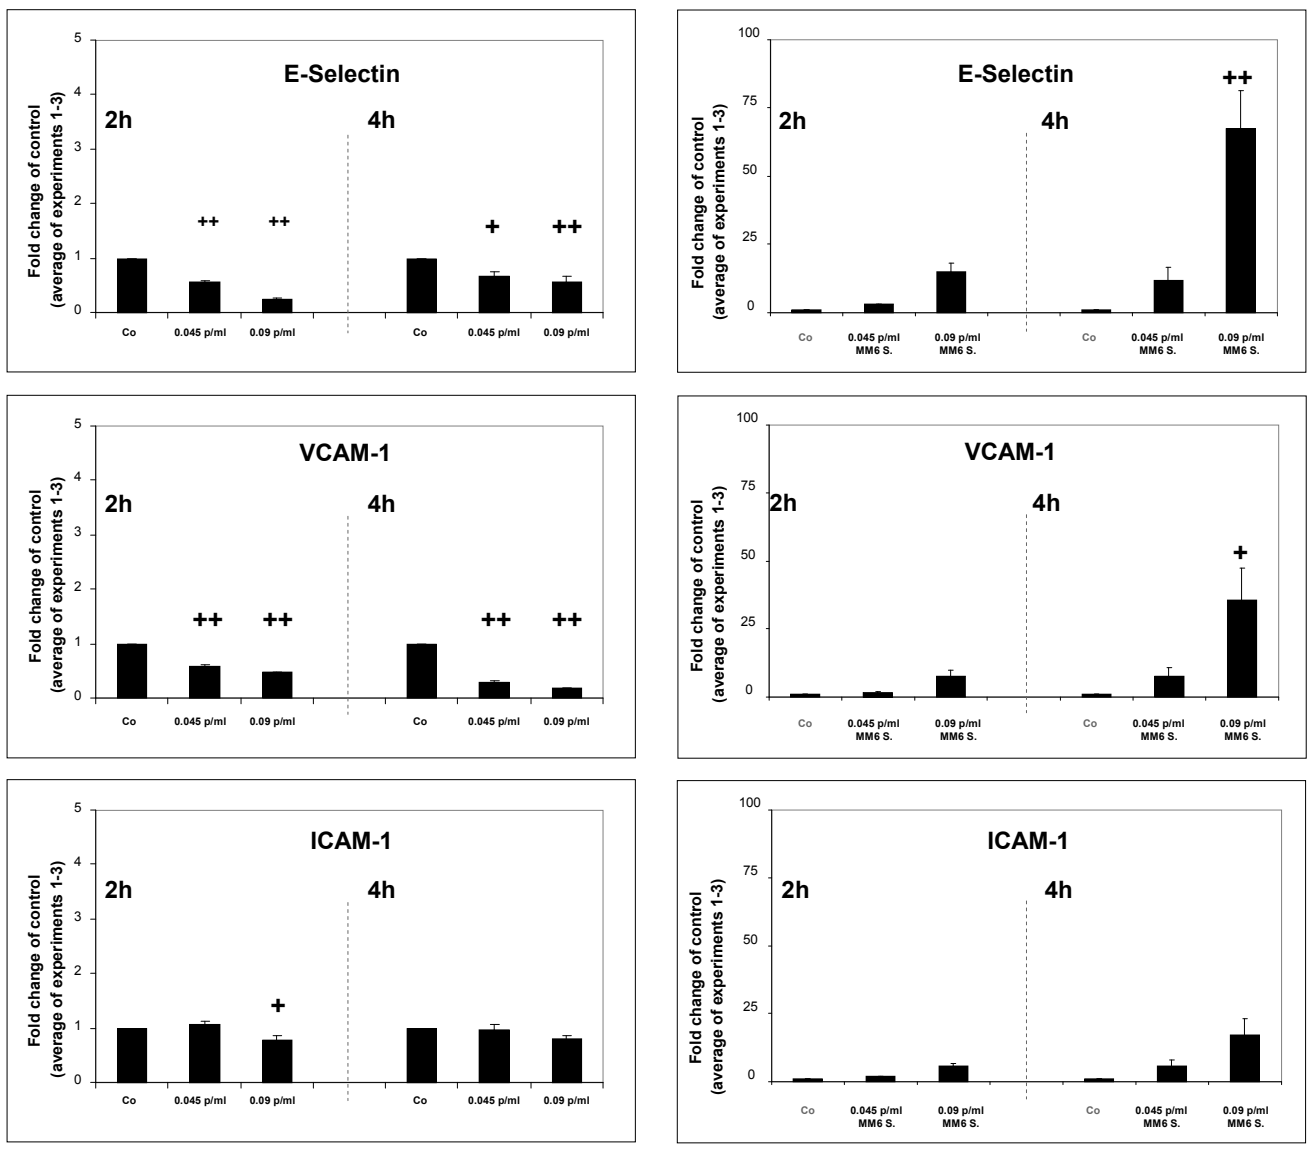
E-Selectin
VCAM-1
ICAM-1
2h
4h
Fold change of control (average of experiments 1-3)
Co
0.045 p/ml
0.09 p/ml
0.045 p/ml MM6 S.
0.09 p/ml MM6 S.

Columns represent the mean of 3 experiments; error bars represent mean ± SE; one-way ANOVA with Dunnett's posthoc test

2) Binding of untreated MM6 cells to CS-exposed HUVECs (indirect treatment) and the impact of blocking antibodies—*in vitro* adhesion assay

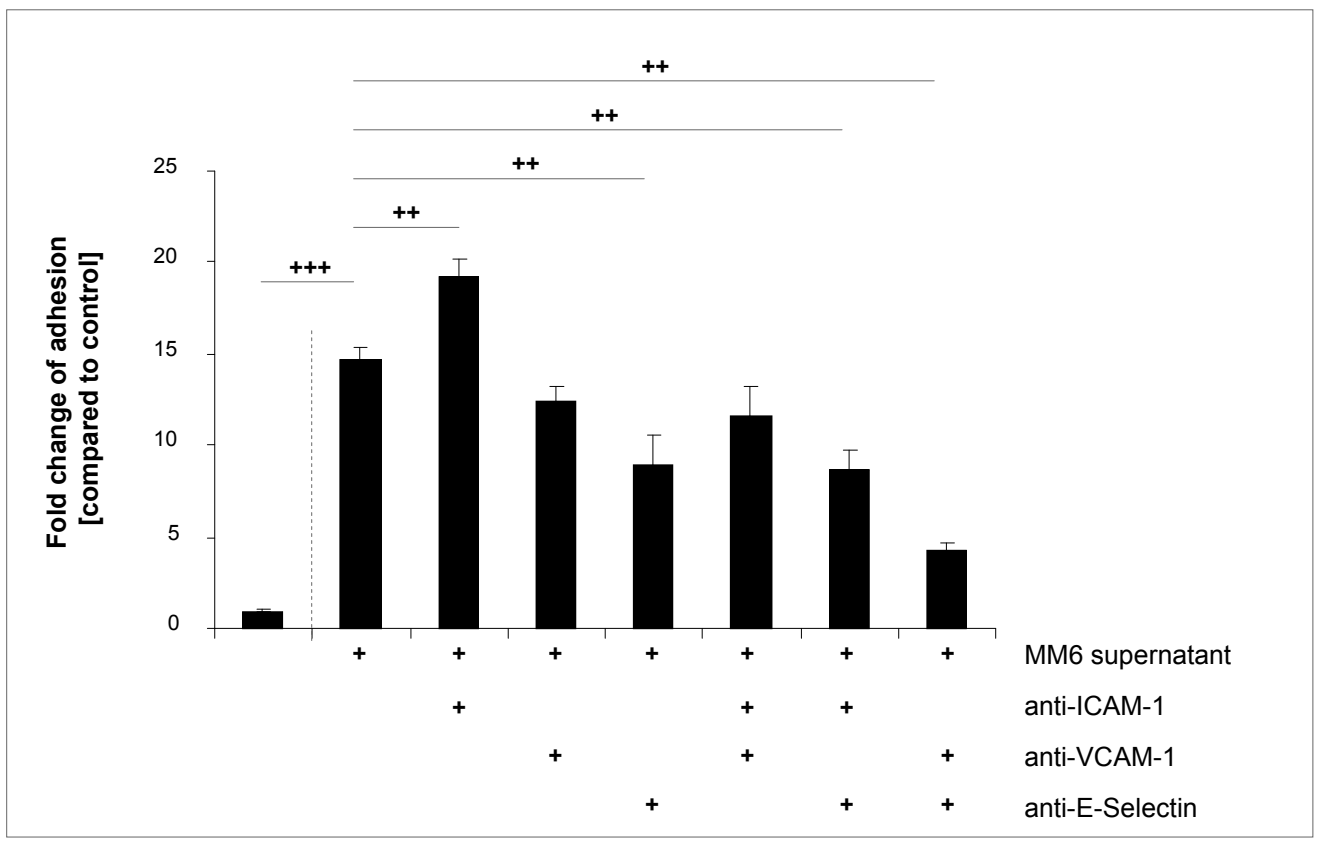
Fold change of adhesion [compared to control]
MM6 supernatant
anti-ICAM-1
anti-VCAM-1
anti-E-Selectin

Columns represent the mean of 3 experiments; error bars represent mean ± SE; t-test (to compare first 2 columns), one-way ANOVA with Dunnett's posthoc test (to compare 2nd column with all others)

3) Binding of CS-exposed MM6 cells (direct treatment, 2h) to untreated HUVECs—*in vitro* adhesion assay

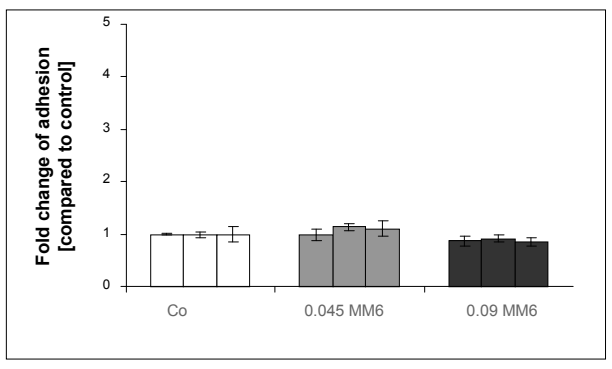
Fold change of adhesion [compared to control]
Co
0.045 MM6
0.09 MM6

Experiments on 3 separate days; error bars represent mean ± SE

4) Up-regulation of Genes in CS-exposed HUVECs (indirect treatment, 4h)—Affymetrix Human Genome U133 2.0

Gene name	Fold change	Remarks
SELE	89.6	**E-Selectin**
CXCL10	74.3	chemokine (C-X-C motif) ligand 10
UBD	67.9	ubiquitin D
VCAM-1	44.6	**vascular cell adhesion molecule 1**
CXCL5	44.3	chemokine (C-X-C motif) ligand 5
ICAM-1	42.3	**intercellular adhesion molecule 1 (CD54)**
CYP1B1	30.3	cytochrome P450, family 1, subfamily B, polypeptide 1
CD69	25.7	CD69 molecule
CCL20	25.3	chemokine (C-C motif) ligand 20
CXCL3	24.4	chemokine (C-X-C motif) ligand 3
NLF1	22.7	nuclear localized factor 1
TRAF1	22.7	TNF receptor-associated factor 1
LTB	22.6	lymphotoxin beta (TNF superfamily, member 3)
SLC7A2	20.4	solute carrier family 7 (cationic amino acid transporter, y+ system), member 2
TNFAIP2	15.7	tumor necrosis factor, alpha-induced protein 2
NPTX1	14.5	neuronal pentraxin I
IL8	13.4	interleukin 8
TNFAIP3	12.9	tumor necrosis factor, alpha-induced protein 3
BIRC3	12.1	baculoviral IAP repeat-containing 3
RND1	10.4	Rho family GTPase 1
TNFAIP3	9.7	tumor necrosis factor, alpha-induced protein 3
NKX3-1	8.8	NK3 transcription factor related, locus 1 (Drosophila)
IL1A	8.4	interleukin 1, alpha
CXCL2	8.1	chemokine (C-X-C motif) ligand 2

Summary and Conclusion

- Direct treatment of HUVECs with CS decreased rather than increased the expression of adhesion molecules (this is contrary to what was seen by Kalra et al. [1994] for cigarette smoke condensate).
- Indirect treatment of HUVECs with CS increased both adhesion molecules expression and the adhesion of MM6 cells to HUVECs *in vitro*.
- In this *in vitro* adhesion assay, the adhesion molecule E-Selectin is a major contributor.
- CS did not increase the adhesiveness of MM6 cells to untreated HUVECs (this is contrary to what was seen by Weber et al. [1996] for isolated monocytes from smokers).

With this *in vitro* adhesion assay, indirect treatment with CS is required to show both increased expression of adhesion molecules in HUVECs and an increased adhesion rate of MM6 cells to HUVECs.

References

a) Libby P. (2002) Inflammation in atherosclerosis. Nature Dec 19-26;420(6917):868-74.

b) Weber C, Erl W, Weber K, Weber PC. (1996) Increased adhesiveness of isolated monocytes to endothelium is prevented by vitamin C intake in smokers. Circulation Apr 15;93(8):1488-92.

c) Kalra VK, Ying Y, Deemer K, Natarajan R, Nadler JL, Coates TD. (1994) Mechanism of cigarette smoke condensate induced adhesion of human monocytes to cultured endothelial cells.J Cell Physiol. Jul;160(1):154-62.

d) Ziegler-Heitbrock HW, Thiel E, Futterer A, Herzog V, Wirtz A, Riethmuller G. (1988) Establishment of a human cell line (Mono Mac 6) with characteristics of mature monocytes. Int J Cancer. 1988 Mar 15;41(3):456-61.

e) University of Kentucky, Kentucky Tobacco Research and Development Center: The Reference Cigarette, Lexington: The University of Kentucky Printing Services, 2003

PMI RESEARCH & DEVELOPMENT

EAS 2010
Hamburg, Germany
20-23 June

Philip Morris International Research & Development, Quai Jeanrenaud 5, 2000 Neuchâtel, Switzerland
T: +41 58 242 21 11, **F**: +41 58 242 28 11, **W**: www.philipmorrisinternational.com