Cigarette Smoke Enhances Foam Cell Formation Independent of Scavenger Receptor Gene Expression

Stolle K[1], Steffen Y[1], Lebrun S[2]
[1]Philip Morris International R&D, Philip Morris Research Laboratories GmbH, Cologne, Germany; [2]Philip Morris International R&D, Philip Morris Products S. A., Neuchâtel, Switzerland

Background and Objective

Epidemiologic research has shown a correlation between cigarette smoking and risk of atherosclerosis[1]. The uptake of modified low density lipoprotein (LDL) is a hallmark of atherosclerotic plaque formation. The modified LDL binds to scavenger receptors, is internalized and leads to foam cell formation, a process considered as a key step in the progression of cigarette-smoke-related atherosclerosis.
In this study, we investigated the impact of cigarette smoke on foam cell formation in macrophages and the underlying mechanisms.

[1] U.S. Department of Health and Human Services. The Health Consequences of Smoking: Cardiovascular Disease. A Report of the Surgeon General Report. Rockville, MD, 2004

Materials and Methods

Monocytes of the human acute monocytic leukemia cell line THP-1 were differentiated to macrophages by phorbol myristate acetate and were treated with mainstream smoke total particulate matter (TPM) from the Reference Cigarette 3R4F (University of Kentucky) for up to 42 hours. The cells were subsequently treated for 4 hours with fluorescence-labelled acetylated LDL (acLDL) for quantification of foam cell formation or with acetylated LDL for RNA analysis. Quality check of foam cell formation was performed by confocal microscopy (data not shown).
For quantitative PCR, TaqMan® Assays (Applied Biosystems) were used. Microarray analysis was performed on Mouse Genome 430 2.0 Arrays (Affymetrix). The quality of all arrays was assessed using the affyQCReport package. Differential gene expression was derived from the GC-RMA background-corrected and normalized signal intensities by averaging biological replicates employing the Bioconductor package. Pathway analysis was performed with IPA5 Ingenuity© software.

Results: Uptake of Acetylated LDL

Uptake of fluorescent-labelled acetylated LDL after 42h: Examples

0 µg TPM 75 µg TPM

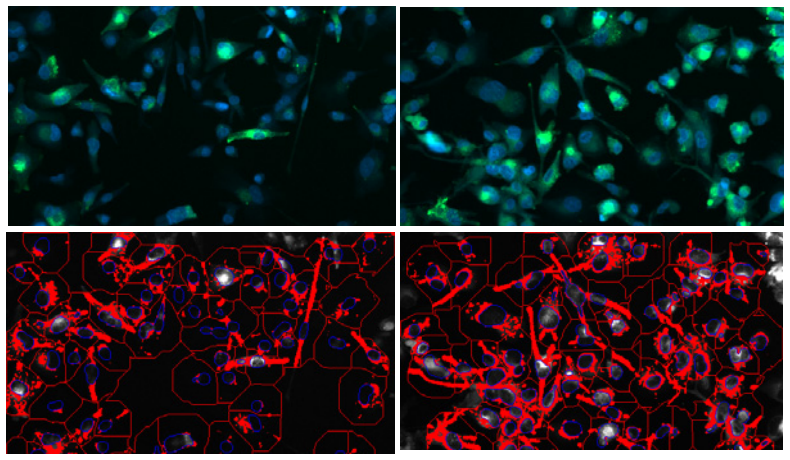

Blue: Nuclei

Green: ALEXA®-acLDL

Blue line: Border of nuclei

Red Area: ALEXA®-acLDL intensity
Red Line: Area for quantification per nucleus

Quantification of fluorescent-labelled acetylated LDL in response to TPM

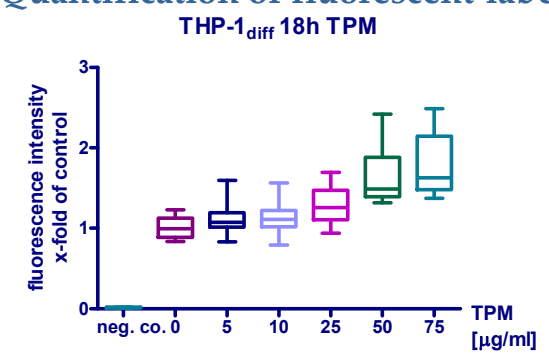
THP-1diff 18h TPM
fluorescence intensity x-fold of control
0
1
2
3
neg. co. 0 5 10 25 50 75
TPM [µg/ml]

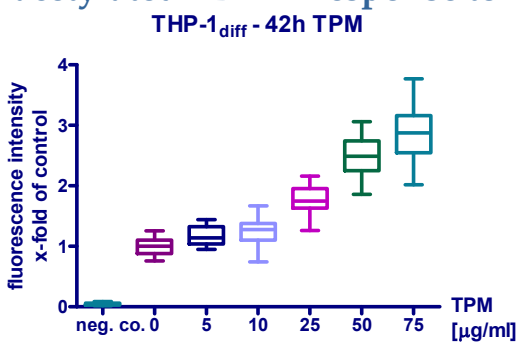
THP-1diff - 42h TPM
fluorescence intensity x-fold of control
0
1
2
3
4
neg. co. 0 5 10 25 50 75
TPM [µg/ml]

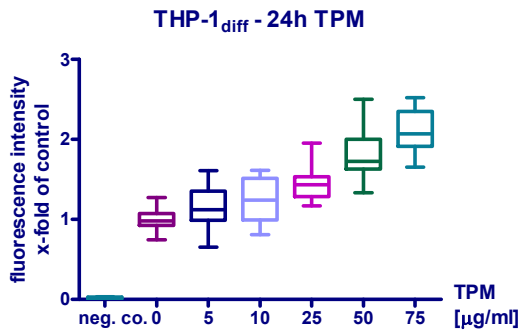
THP-1diff - 24h TPM
fluorescence intensity x-fold of control
0
1
2
3
neg. co. 0 5 10 25 50 75
TPM [µg/ml]

Neg.co.: Cells without acLDL or TPM
Shown are median (line), 1st and 3rd quartiles (box) and minimum and maximum values (whiskers)

Results: Gene Expression and Pathway Analysis

Gene expression of scavenger receptors in response to TPM

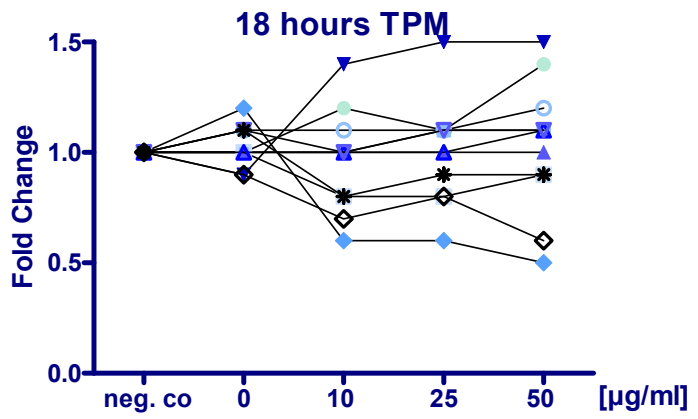
18 hours TPM
Fold Change
0.0
0.5
1.0
1.5
neg. co 0 10 25 50 [µg/ml]

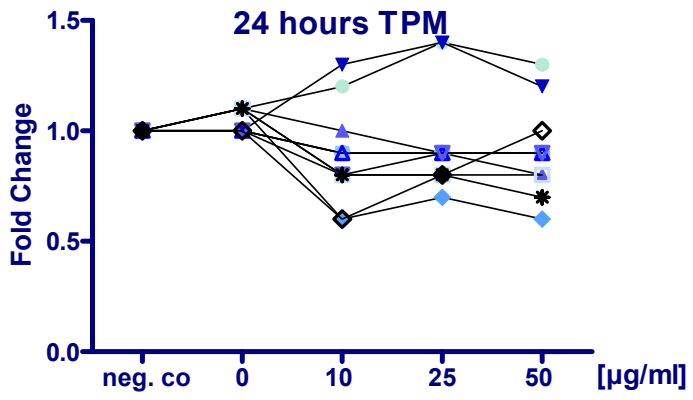
24 hours TPM
Fold Change
0.0
0.5
1.0
1.5
neg. co 0 10 25 50 [µg/ml]

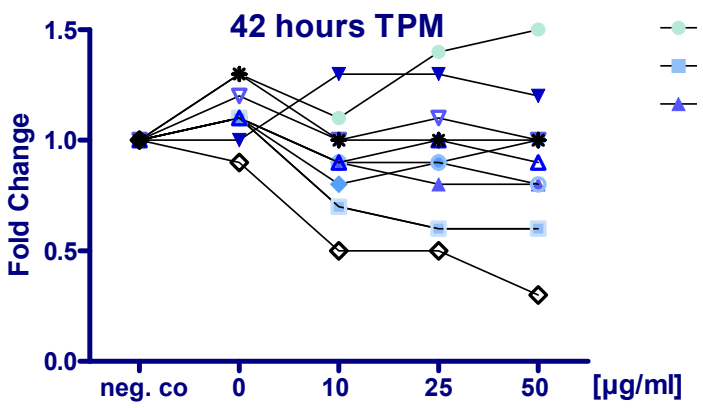
42 hours TPM
Fold Change
0.0
0.5
1.0
1.5
neg. co 0 10 25 50 [µg/ml]

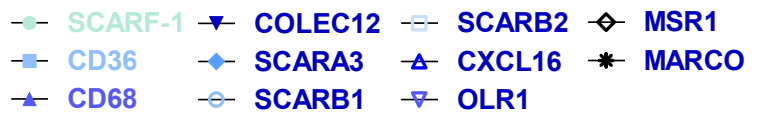
SCARF-1
CD36
CD68
COLEC12
SCARA3
SCARB1
SCARB2
CXCL16
OLR1
MSR1
MARCO

Shown is the average of 3 experiments (relative quantification value as fold change)

Class	Scavenger Receptor
A	MSR1 (macrophage scavenger receptor 1), MARCO (macrophage receptor with collagenous structure), SCARA3 (scavenger receptor class A, member 3), and COLEC12 (collectin sub-family member 12)
B	CD36, SCARB1, and SCARB2 (CD36 molecule (thrombospondin receptor, scavenger receptor class B, member 1 and 2)
D	CD68 (Macrophage antigen CD68 (macrosialin))
E	OLR1 (oxidized low density lipoprotein (lectin-like) receptor 1)
F	SCARF1 and SCARF2 (scavenger receptor class F, member 1 and 2
G	SR-PSOX (scavenger receptor for phosphatidylserine and oxidized low density lipoprotein)

Identification of pathways (underlying mechanisms)

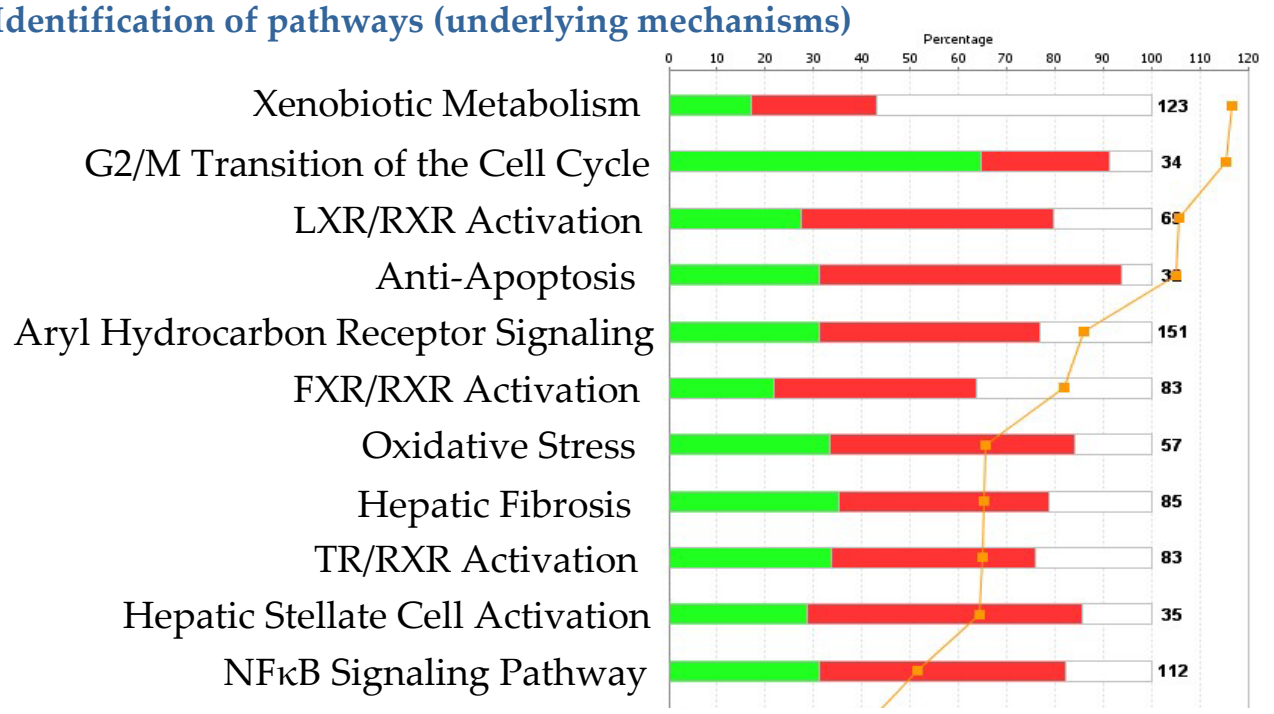
Percentage
0 10 20 30 40 50 60 70 80 90 100 110 120
Xenobiotic Metabolism
G2/M Transition of the Cell Cycle
LXR/RXR Activation
Anti-Apoptosis
Aryl Hydrocarbon Receptor Signaling
FXR/RXR Activation
Oxidative Stress
Hepatic Fibrosis
TR/RXR Activation
Hepatic Stellate Cell Activation
NFκB Signaling Pathway
123
34
151
83
57
85
83
35
112

-log(p-value)
Down-regulated
Up-regulated

Thyroid Hormone Receptor (TR)
Retinoid X Receptor (RXR)
Liver X Receptor (LXR)

Summary

Foam cell formation of THP-1 macrophages is enhanced dose-dependently by treatment with mainstream smoke total particulate matter (TPM), independent of differential gene expression of scavenger receptors. This was shown by:

- The uptake of fluorescence-labelled LDL in THP-1 macrophages treated with TPM was enhanced dose-dependently.
- Exposure to TPM did not affect the gene expression of scavenger receptors.
- Genes involved in oxidative stress, NFkB signaling and other pathways are affected by TPM.

Conclusion

Scavenger receptors are reported to be involved in foam cell formation. Our results show that no *newly* synthesized scavenger receptors are required for the acceleration of foam cell formation by TPM. However, the mode of action remains to be elucidated, i.e., whether a facilitated internalization of scavenger receptors or other pathways are involved.

PMI RESEARCH & DEVELOPMENT

Philip Morris International Research & Development, Quai Jeanrenaud 5, 2000 Neuchâtel, Switzerland
T: +41 58 242 21 11, **F**: +41 58 242 28 11, **W**: www.philipmorrisinternational.com