Contribution of single smoke constituents to the mutagenic activity of the gas/vapor phase of cigarette mainstream smoke

S. Weber[1], T.J. Meisgen[1], J. Diekmann[1], A. Wittig[1], K. Rustemeier[2]
[1]Philip Morris Research and Development, Philip Morris Research Laboratories GmbH, Cologne, Germany; [2]Rustemeier Consulting, Meckenheim, Germany

Introduction and Objective

The gas/vapor phase (GVP) of cigarette mainstream smoke contains a number of single constituents that have been shown to be mutagenic in the *Salmonella* reverse mutation assay (e.g., Rodgman and Green, 2002). In the microsuspension version of the *Salmonella* reverse mutation assay the gas/vapor phase of cigarette smoke has been shown to be mutagenic, with acrolein appearing to be the major contributor to GVP mutagenicity (Tewes et al., 2004).

To further investigate the contribution of single smoke constituents to the mutagenic activity of the GVP of cigarette mainstream smoke, we first screened the water-soluble fraction of the GVP of mainstream smoke from the Reference Cigarette 2R4F for smoke constituents and then selected constituents for mutagenicity screening.

Methods

Cigarettes and GVP collection

- University of Kentucky Reference Cigarette 2R4F (total particulate matter [TPM] yield ~12 mg/cig.).
- Cigarettes (20 per batch) were smoked on a 20-port Borgwaldt smoking machine in basic conformity with ISO standards (1991).
- TPM was filtered out by passing the smoke through glass fiber filters.
- The remaining GVP was bubbled through 50 ml ice-cold water (for constituent screening) or phosphate-buffered saline (PBS) (for mutagenicity testing) in a gas wash bottle, and the water-soluble portion was trapped.

Gas chromatography/mass spectrometry (GC/MS) fingerprinting

- Internal standard (d6-phenol) was added to an aliquot of the GVP sample.

<u>Gas chromatography</u>

- Hewlett Packard 6890 equipped with a cool on-column injector.
- Chromatography carried out on a 30 m capillary DB-FFAP column (0.25 mm x 0.25 µm) with a gas flow of 1ml/min.
- Injector temperature ramped to follow oven temperature.
- Temperature program: 30°C 5°/min to 40°C 10°/min to 200°C and 5°/min to 250°C for 10 min).

<u>Mass spectrometry</u>

- Hewlett Packard 5973 MSD operating in EI ionization mode.
- Ion source temperature 250°C and quadrupole set to 150°C.

<u>Peak identification</u>

- Mass spectral comparison with standard mass spectral libraries NIST02 and WILEY7N.
- Semiquantitative yields derived by relating the combined areas of target- and qualifier-ions for each compound to the combined areas of target- and qualifier-ions for the internal standard d6-phenol.

Mutagenicity testing

- Microsuspension version of the *Salmonella* reverse mutation assay according to Kado et al, 1983.
- *Salmonella typhimurium* his- tester strain TA100, cultivated at 36°C for approximately 11 h.
- TA100 without exogenous metabolic promutagen activation.
- GVP and pure substances were tested.
- GVP evaluation was based on the amount of TPM trapped (TPM equivalents).
- Pure substances (commercially obtained; purity ≥95% [exception: formaldehyde, purity ~37%]) were dissolved in PBS.
- At least 4 doses assayed per sample; doses selected to cover the increase in mutagenicity.
- Bacteria exposed for 120 min (pre-incubation), 3 cultures per dose.

Results

GVP screening for smoke constituents

GC/MS fingerprinting identified <u>65 active smoke constituents</u> from <u>13 chemical classes</u>, with the class of aldehydes having the highest yields.

Selection of smoke constituents for mutagenicity screening

The 10 single constituents with the highest yields plus 5 other constituents reported to be mutagenic, representing 6 chemical classes, were assayed as pure substances for mutagenic activity. These 15 constituents represent approximately 87% of the total yield of the 65 GVP constituents.

Of the 15 constituents tested, 6 were found to be mutagenic with a response at least 2-fold higher than the spontaneous reversion, all observed in the applied dose range. The 6 mutagenic constituents represent approximately 15% of the total yield of the 65 GVP constituents.

Substance (chemical class)	Structure	Yield (µg/cig.)	% of chemical class represented[a]	Mutagenic (maximum -fold induction)
Constituents with highest Yields				
acetaldehyde	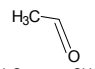	490	77% of aldehydes	no
acetone	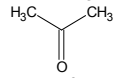	415	75% of ketones	no
2,3-butanedione	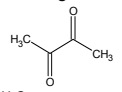	103	88% of diketones	**yes (2.2)**
2-butanone	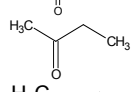	89.5	16% of ketones	no
acetonitrile	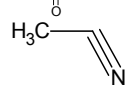	64.4	61% of nitriles	no
acrolein	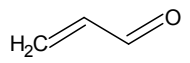	63.2	10% of aldehydes	**yes (5.8)**
propionaldehyde	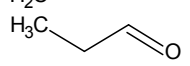	29.3	5% of aldeyhdes	no
methyl vinyl ketone	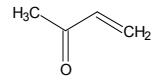	28.7	5% of ketones	**yes (4.3)**
crotonaldehyde	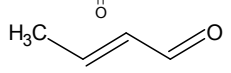	18.2	3% of aldeyhdes	**yes (9.7)**
Toluene	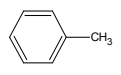	14.1	50% of aromatic hydrocarbons	no
Additional Mutagenic Constituents				
Methacrolein	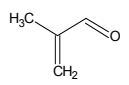	9.78	2% of aldehydes	**yes (4.7)**
formaldehyde (detected by HPLC)	$H_2C{=}O$	7.93	1% of aldehydes	**yes (7.7)**
benzene	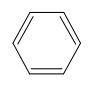	5.96	21% of aromatic hydrocarbons	no
Pyridine	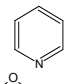	2.75	23% of n-heterocycles	no
2-furaldehyde	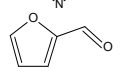	0.71	0.1% of aldehydes	no

[a] % of chemical class represents the proportion of the yield of each chemical class identified in the GC/MS-Fingerprint. Chemical classes that were detected, but not represented in the mutagenicity screening were aliphatic hydrocarbons, halogenated compounds, sulfur compounds, esters, and ethers, o-heterocycles, alcohols, and short chain and aromatic acids.

Contribution of single constituents to GVP mutagenicity

- **Based on equal effect concentration ratios from the non-linear dose response curves, it can be estimated that acrolein contributes 60%, crotonaldehyde 13%, formaldehyde 3%, and all others less than 3% to the mutagenic activity of the GVP.**

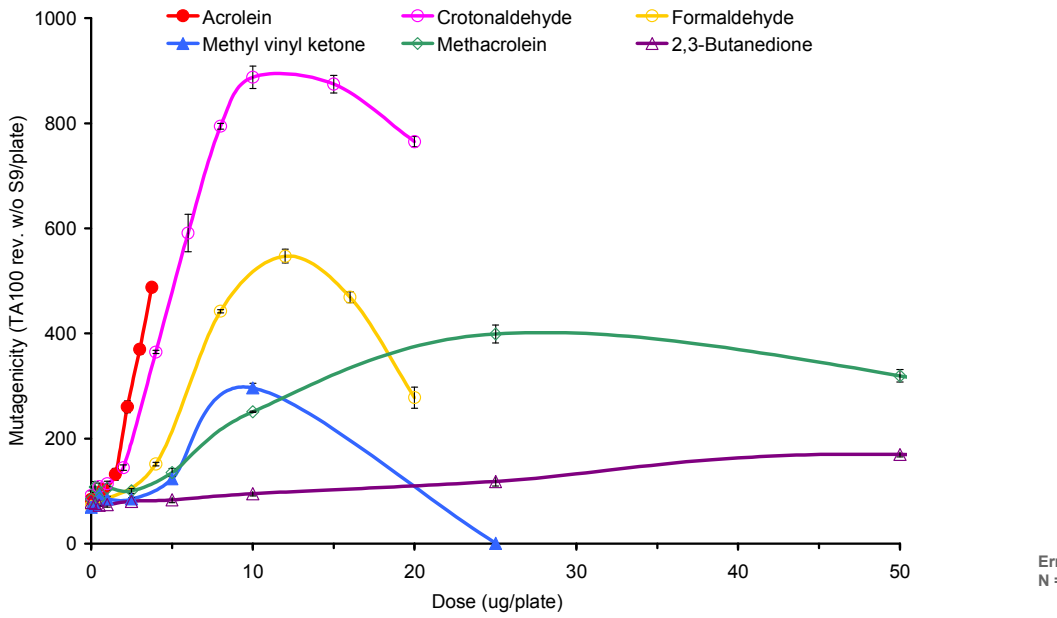
Acrolein
Crotonaldehyde
Formaldehyde
Methyl vinyl ketone
Methacrolein
2,3-Butanedione
Mutagenicity (TA100 rev. w/o S9/plate)
1000
800
600
400
200
0
0
10
20
30
40
50
Dose (ug/plate)

Error bars represent mean ± SD from N = 3 plates

Contribution of carbonyls to GVP mutagenicity

The six active constituents were combined as pure substances in the same concentration ratios as determined in the GVP solution, which was tested in parallel.

- **60% of the mutagenicity of the GVP can be attributed to the mutagenicity of 3 aldehydes: acrolein, crotonaldehyde, and formaldehyde (MIX 1).**
- **The addition of methyl vinyl ketone, methacrolein, and 2,3-butanedione (MIX 2) did not contribute remarkably to the mutagenicity.**

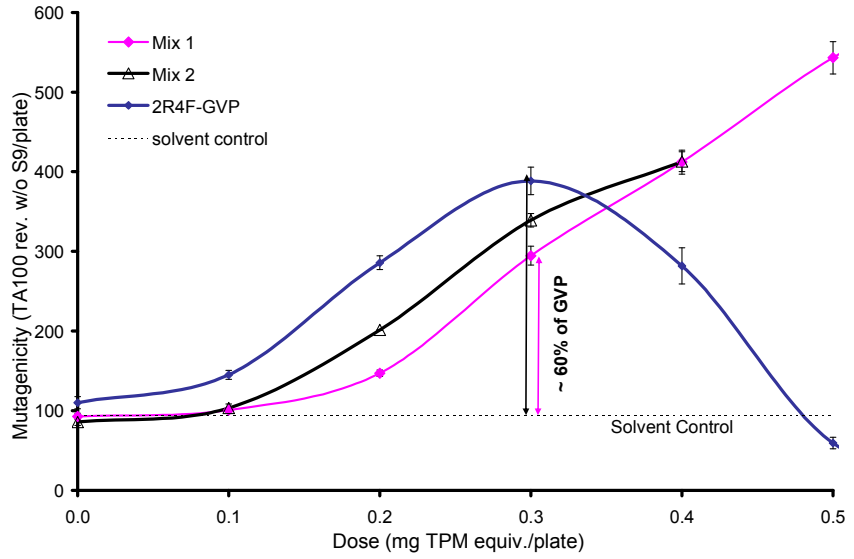
Mix 1
Mix 2
2R4F-GVP
solvent control
Mutagenicity (TA100 rev. w/o S9/plate)
600
500
400
300
200
100
0
0.0
0.1
0.2
0.3
0.4
0.5
Dose (mg TPM equiv./plate)
~ 60% of GVP
Solvent Control

MIX 1:
- acrolein: ~5.0 µg/mg TPM equivalents
- crotonaldehyde: ~1.1 µg/mg TPM equivalents
- formaldehyde: ~0.7 µg/mg TPM equivalents

MIX 2:
- aldehyde Mix 1 plus
- methacrolein: ~1.1 µg/mg TPM equivalents
- methyl vinyl ketone: ~2.5 µg/mg TPM equivalents
- 2,3-butanedione: ~8.2 µg/mg TPM equivalents

Error bars represent mean ± SD from N = 3 plates

Summary

Six constituents were positive for mutagenic activity in the microsuspension version of the *Salmonella* reverse mutation assay: acrolein, crotonaldehyde, formaldehyde, methyl vinyl ketone, methacrolein, and 2,3-butanedione.

Acrolein is the major contributor to the overall mutagenicity, followed by crotonaldehyde and formaldehyde. Together these three aldehydes account for approximately 60% of the mutagenicity of GVP from the 2R4F. The contribution of the other three constituents that were positive for mutagenic activity (methyl vinyl ketone, methacrolein, and 2,3-butanedione), was minimal.

References

- Kado et al., Mutat. Res. 121: 25-32 (1983)
- International Organization for Standardization: International Standard ISO 3308, Cigarettes - Routine analytical cigarette-smoking machine - Definitions and standard conditions, 3rd ed., 1991
- Rodgman and Green, CORESTA SYMP., 2002, pp. 2-52 (2002)
- Tewes et al., Poster, Society of Toxicology Meeting (2004)

PMI RESEARCH & DEVELOPMENT

Society of Toxicology
49th Annual Meeting & ToxExpo™
Salt Lake City, UTAH
7-11 March 2010

Philip Morris International Research & Development, Quai Jeanrenaud 5, 2000 Neuchâtel, Switzerland
T: +41 58 242 21 11, **F**: +41 58 242 28 11, **W**: www.philipmorrisinternational.com