Cigarette-smoke-induced cellular transformation *in vitro* using the Bhas 42 cell transformation assay

D. Weisensee and J. Schüller
Philip Morris Research and Development, Philip Morris Research Laboratories GmbH, Cologne, Germany

Introduction and Objective

In vitro cell transformation assays detect transformed cells that have acquired the characteristics of malignant cells and thus mimic one stage of the *in vivo* multi-step carcinogenesis model. These assays have been proposed as surrogate models for predicting the non-genotoxic carcinogenic potential of chemicals. A short-term cell transformation assay using *v-Ha-ras*-transfected Balb/c 3T3 cells (Bhas 42 cells), the Bhas assay, which has not been used with cigarette smoke before, is capable of detecting initiating and promoting activities of chemical carcinogens (Asada et. al, 2005 and Ohmori et. al, 2005). As the particulate phase of cigarette smoke (total particulate matter, or TPM) is known to induce tumors *in vivo* in the mouse skin painting assay, we investigated the responsiveness of the Bhas assay to form morphologically transformed foci *in vitro* when repeatedly challenged with TPM from a standard reference cigarette.

The objective of the present study was to induce morphologically transformed foci by the repeated application of total particulate matter (TPM) derived from the Reference Cigarette 3R4F in a dose-dependent manner.

Materials and Methods

Cells and Cell Culture Conditions

Bhas 42 cells are *v-Ha-ras*-transfected Balb/c 3T3 clone A31-1-1 mouse fibroblasts isolated and characterized by Sasaki et al. (1988). Bhas 42 cells were obtained from Hatano Research Institute (HRI), Hatano, Japan, and were used for the morphological transformation assay at Harlan CCR, Rossdorf, Germany, where this study was conducted. Stock cultures were propagated at 37 °C in 75 cm^2 plastic flasks and about 2.5 x 10^5 cells were put into two flasks with 15 mL each of M10F* medium.

Positive control: 12-O-tetradecanoylphorbol-13-acetate (TPA)
Negative control: dimethyl sulfoxide (DMSO)

* M10F: minimal essential medium supplemented with 10% fetal bovine serum

Cell Growth Assay

- Thaw cells and pre-culture for 3 to 4 days in M10F culture medium then replace culture medium with DF5F* for 3 days.
- At 70% confluence: trypsinize and seed in 6-well microtiter plates (0.7 x 10^4/mL) for 4 days.
- Replace culture medium with fresh medium containing test substance for 3 days.
- Fix cells with methanol, stain with crystal violet, air-dry, and count.

*DF5F: Dulbecco's modified Eagle's medium/F12 supplemented with 5% fetal bovine serum

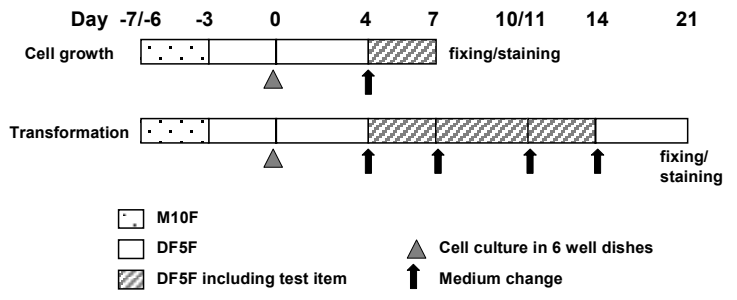
Day -7/-6 -3 0 4 7 10/11 14 21
Cell growth
fixing/staining
Transformation
fixing/ staining
M10F
DF5F
DF5F including test item
Cell culture in 6 well dishes
Medium change

Morphological Transformation (Bhas Assay)

- Thaw cells and pre-culture for 3 to 4 days in M10F culture medium then replace culture medium with DF5F* for 3 days.
- At 70% confluence trypsinize and seed in 6-well microtiter plates (0.7 x 10^4/mL) for 4 days.
- Replace culture medium with fresh medium containing test substance for 3-4 days (3 times).
- Replace culture medium with fresh medium without test substance for 7 days (recovery).
- Fix cells with methanol, stain with crystal violet, air-dry, and count type III foci.

Only type III foci are considered tumorigenic because they are the only that induce neoplasic transformation in nude mice. Type III foci are basophilic and densely multilayered with spindle-shaped cells that are randomly oriented at the focus edge and invade the monolayer.

Generation of Total Particulate Matter (TPM)

The Reference Cigarette 3R4F (University of Kentucky) was smoked on a Borgwaldt 20-port smoking machine (16 cigarettes, 8 puffs/cigarette, 35 mL puff volume). TPM was collected on a glass fiber filters. The TPM from one filter was extracted with 3 mL DMSO to generate a final concentration of ~ 50 mg TPM/mL.

Results

Relative Cell Growth

- Single values after 3-day exposure to TPM.

	Concentration	Optical density			Mean	SD	Relative growth[a]
	µg TPM/mL	well 1	well 2	well 3			%
Neg. control[b]		0.487	0.496	0.480	0.488	0.008	
Solv. control[c]		0.440	0.477	0.484	0.467	0.024	100.0
Pos. control[d]		0.461	0.550	0.504	0.505	0.045	108.1
Test item	5.0	0.440	0.466	0.519	0.475	0.040	101.7
Test item	10.0	0.373	0.500	0.418	0.431	0.064	92.2
Test item	20.0	0.528	0.375	0.493	0.466	0.080	99.6
Test item	30.0	0.493	0.426	0.431	0.450	0.037	96.4
Test item	40.0	0.440	0.438	0.477	0.452	0.022	96.7
Test item	50.0	0.490	0.438	0.442	0.457	0.029	97.8
Test item	60.0	0.439	0.485	0.436	0.454	0.027	97.1
Test item	70.0	0.370	0.404	0.402	0.392	0.019	84.0
Test item	80.0	0.409	0.432	0.441	0.428	0.017	91.5
Test item	100.0	0.189	0.228	0.219	0.212	0.020	45.4
Test item	102.0	0.105	0.106	0.094	0.102	0.007	21.8

[a] Compared to solvent control
[b] Complete culture medium
[c] DMSO 0.5% (v/v)
[d] TPA 50 ng/mL

- Cytotoxicity (indicated by a 50% reduction in cell growth relative to control) was observed at 100 µg/mL and above.

Examples of Type III Foci *

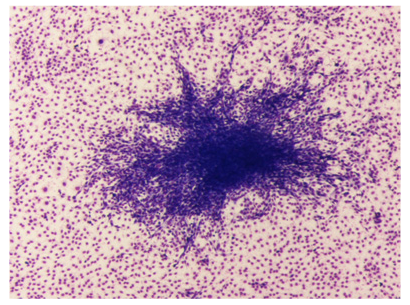

- Type III focus and surrounding intact cell monolayer with normal morphology

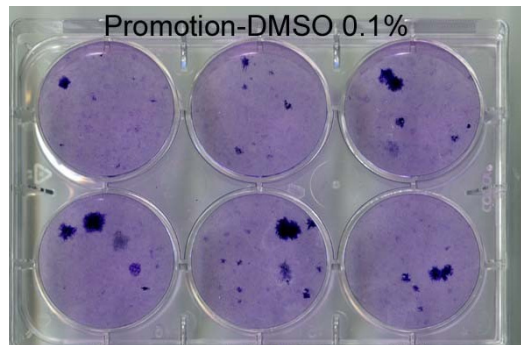
Promotion-DMSO 0.1%

- Type III foci in DMSO negative control

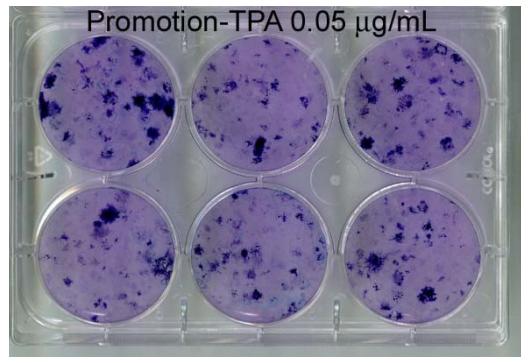
Promotion-TPA 0.05 µg/mL

- Type III foci in TPA positive control

* The images were kindly provided by Dr. A. Poth (Harlan CCR, Rossdorf, Germany)

Morphological Transformation

The test item is considered positive/transforming if the following criterion is met: the number of morphologically transformed foci is statistically significantly increased at least at two concentrations compared to the concurrent solvent control.

Test item concentration	Relative cell growth[a]	Number of transformed foci type III	
µg TPM/mL	%	mean	sum
Exposure period 10 days (repeated application)			
Negative control[b]		1.8	11
Solvent control[c]	100.0	2.5	15
Positive control[d]	108.1	11.8*	71
5.0	101.7	10.5*	63
10.0	92.2	21.8*	131
20.0	99.6	39.0*	234
30.0	96.4	40.8*	245
40.0	96.7	35.8*	215
50.0	97.8	27.2*	163
60.0	97.1	20.7*	124
70.0	84.0	3.8	23
80.0	91.5	0.0	0
100.0	45.4	0.0	0
120.0	21.8	0.0	0

[a] Treatment of cultures in the cell growth assay was 3 days
[b] Complete culture medium
[c] DMSO 0.5% (v/v)
[d] TPA 50 ng/mL
* Statistically significant compared to control; confirmed by the one-sided Dunnett test ($p < 0.05$) using SigmaSTAT®2.0

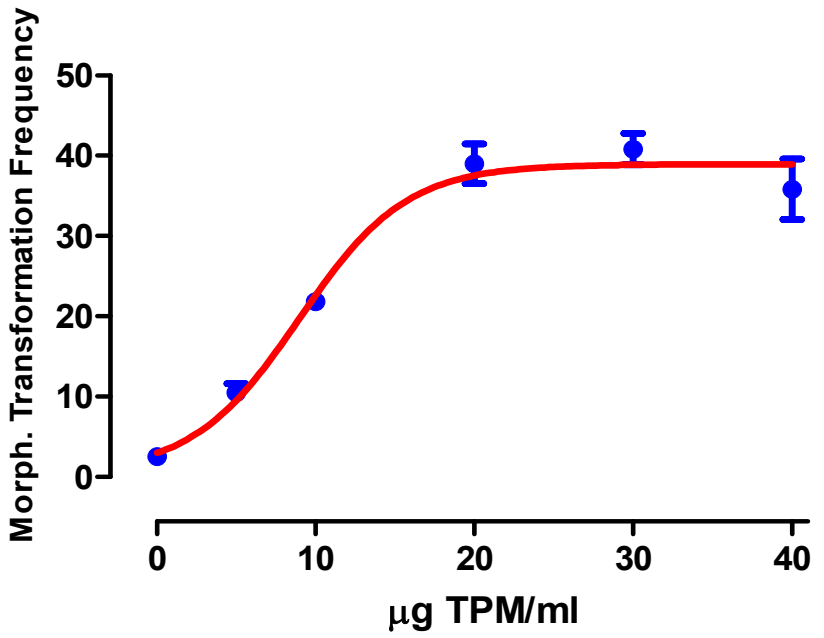
Morph. Transformation Frequency
50
40
30
20
10
0
0 10 20 30 40
µg TPM/ml

Error bars represent the mean ± SD

- TPA induced a statistically significant increase ($p < 0.05$) in morphologically transformed foci.
- Statistically significant increases in the frequency of morphologically transformed foci (~16-fold) were seen at TPM concentrations 5 to 60 µg/mL.
- The decrease or lack of morphologically transformed foci at TPM concentrations 70 to 120 µg/mL is due to cytotoxic effects after repeated application of TPM for 10 days.

Conclusion

Under the experimental conditions reported, TPM from the Reference Cigarette 3R4F induced a dose-dependent increase of morphologically transformed foci after repeated application for 10 days. Cigarette smoke, which is known to be tumorigenic in the *in vivo* mouse skin painting assay, is shown to induce morphological transformation *in vitro* in the Bhas 42 cell transformation assay.

This novel *in vitro* assay using Bhas 42 cells, which are regarded as initiated in the two-stage paradigm of carcinogenesis, is able to detect cell transformation induced by cigarette smoke condensate in a dose-dependent manner with a high dynamic range (~16-fold increase).

References

Asada, S., Sasaki, K., Tanaka, N., et. al (2005)
Detection of initiating as well as promoting activity of chemicals by a novel cell transformation assay using v-Ha-ras-transfected BALB/c 3T3 cells (Bhas 42 cells). *Mutat. Res.* **588**, 7-21

Ohmori, K., Umeda, M., Tanaka, N., et. al (2005)
An Inter-laboratory Collaborative Study by the Non-Genotoxic Carcinogen Study Group in Japan, on a Cell Transformation Assay for Tumour Promoters Using Bhas 42 cells. *ATLA* **33**, 619-639

Sasaki, K., Mizusawa, H., and Ishidate, M. (1988)
Isolation and Characterization of *ras*-Transfected BALB/c 3T3 Clone Showing Morphological Transformation by 12-*O*-Tetradecanoyl-phorbol-13-acetate. *Jpn. J. Cancer Res. (Gann)* **79**, 921-930

PMI RESEARCH & DEVELOPMENT

Society of Toxicology
49th Annual Meeting & ToxExpo™
Salt Lake City, UTAH
7-11 March 2010

Philip Morris International Research & Development, Quai Jeanrenaud 5, 2000 Neuchâtel, Switzerland
T: +41 58 242 21 11, **F**: +41 58 242 28 11, **W**: www.philipmorrisinternational.com