Cigarette-smoke-dependent changes in the respiratory and cardiovascular system of spontaneously hypertensive (SH) rats and the effect of smoking cessation

A. Berges[1], B. Friedrichs[2], I. Gallitz[2], S. Quadt-Humme[2], Y. Steffen[2], K. Stolle[2], S. Lebrun[3], and **M. Lietz**[2]

[1 & 2] Philip Morris Research Laboratories bvba, Leuven, Belgium and GmbH, Cologne, Germany; [3]Philip Morris Products SA, Neuchâtel, Switzerland

SOT sponsor: T. Mueller

Abstract #187

Introduction and Objective

Based on its propensity to develop spontaneously hypertension-related cardiovascular diseases,[1] the spontaneously hypertensive (SH) rat is considered a relevant model to investigate cigarette smoke effects on cardiac function and hypertrophy.[2-3]

The SH rat displays phenotypes such as systemic inflammation, hypercoagulation, oxidative stress, and suppressed immune function, which are also apparent in COPD patients. A cigarette-smoke-induced inflammatory response in the airways and lung parenchyma is believed to be the major cause of chronic obstructive pulmonary disease (COPD) in Smokers.[4]

The **objective** of the study was three-fold:

1. To confirm the effects of cigarette smoke on cardiac hypertrophy status seen in a previous study (see Abstract 912, SOT 2008)
2. To investigate the effects of cigarette smoke on lung inflammation
3. To investigate the reversibility of these effects after smoking cessation

Materials and Methods

Animal treatment

The *in vivo* study was performed at Philip Morris Research Laboratories bvba, Leuven, Belgium. Care and use of the animals was in accordance with the American Association for Laboratory Animal Science Policy (1996). All animal experiments were approved by the Institutional Animal Care and Use Committee (IACUC).

Exposure conditions

Adult male SH rats were nose-only exposed to filtered, conditioned air (sham) or to mainstream smoke (MS) from the Reference Cigarette 3R4F at a total particulate matter (TPM) concentration of 900 µg/day; 5 days/week for 90 days.

Subsets of SH rats were exposed for 30 days to cigarette smoke followed by 60 days of SHAM exposure (SC-1) or for 60 days to cigarette smoke followed by 30 days of SHAM exposure (SC-2).

Biological endpoints

- Body weight (weekly)
- Total nicotine metabolites in urine (overnight), immediately prior to smoke cessation and/or dissection
- Cardiac hypertrophy, i.e., heart weight vs. body weight or tibia length
- Determination of total free lung cells in bronchoalveolar lavage fluid (BALF), lung cell differentiation and alveolar macrophage activation markers (flow cytometry)
- Quantification of COPD-relevant mediators in BALF (ELISA):
 - --*imbalance protease/antiprotease*: MMP-1, MMP-8, MMP-2, MMP-12, TIMP-1, elastase-2, α1-antitrypsin, perforin 1
 - --*chemokines and cytokines*: MCP-1, CXCL-9, CXCL-11, IFN-α, IFN-γ, TGF- α, TGF-β1, TGF-β2

(box plots: median, lower and upper quartile, min and max)

References

1. Doggrell SA, Brown L. Rat models of hypertension, cardiac hypertrophy and failure. *Cardiovasc Res.* 1998 Jul;39(1):89-105
2. Meurrens K, Ruf S, Ross G, Schleef R, von Holt K, Schlüter KD. Smoking accelerates the progression of hypertension-induced myocardial hypertrophy to heart failure in spontaneously hypertensive rats. *Cardiovasc Res*. 2007 Nov 1;76(2):311-22
3. Schleef R, Meurrens K, Berges A, Stolle K, Eggert C, Diehl S, Wallerath T, Lietz M. Mainstream cigarette smoke (MS) affects the cardiovascular system of spontaneously hypertensive rats (SHR). Abstract 912 SOT 2008
4. Yu B, Kodavanti UP, Takeuchi M, Witschi H, Pinkerton KE. Acute tobacco smoke-induced airways inflammation in spontaneously hypertensive rats. *Inhal Toxicol*. 2008 May;20(7):623-33.

Results

Body Weight

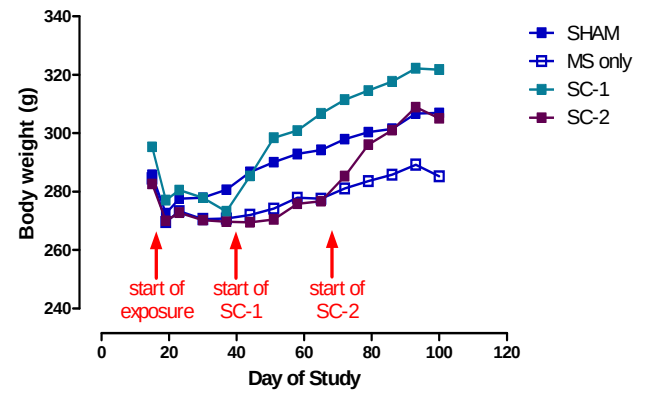
Body weight (g)
Day of Study
SHAM
MS only
SC-1
SC-2
start of exposure
start of SC-1
start of SC-2

	BW at start of exposure (g)	BW at dissection (g)	Absolute difference (g)	Relative difference (%)
SHAM	285.7	306.9	21.2	7.4
MS only	284.5	285.2	0.7	0.2
SC-1	295.3	321.8	26.5	9.0
SC-2	282.6	305.1	22.5	8.0

Nicotine Metabolites in Urine

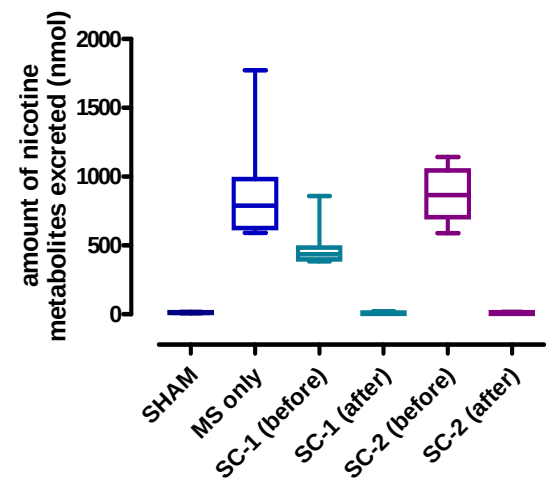
amount of nicotine metabolites excreted (nmol)
SHAM
MS only
SC-1 (before)
SC-1 (after)
SC-2 (before)
SC-2 (after)

Cardiac Hypertrophy (3m exposure

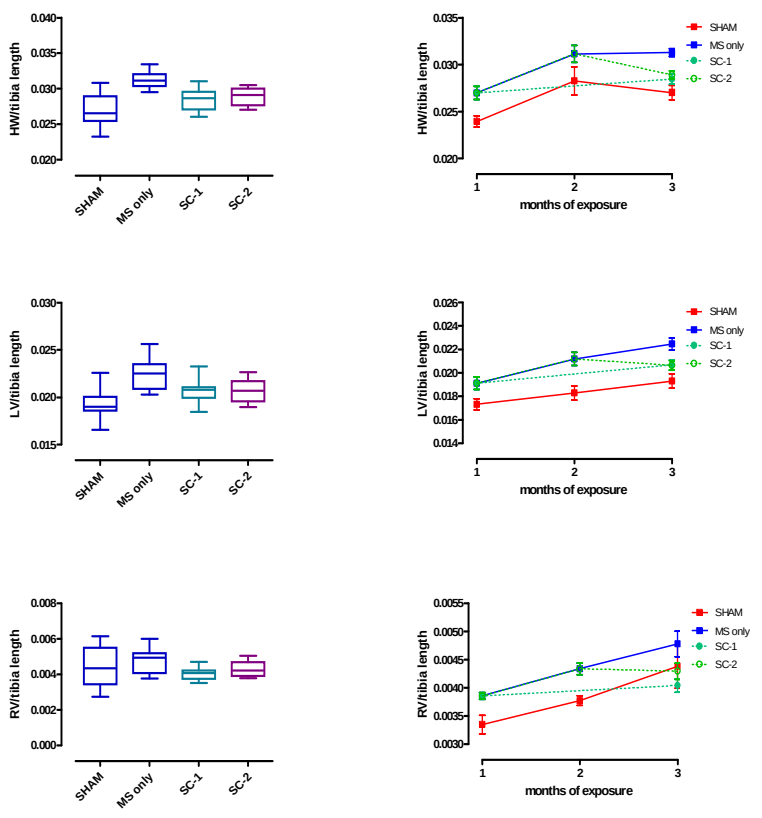
HW/tibia length
LV/tibia length
RV/tibia length
months of exposure
SHAM
MS only
SC-1
SC-2

HW: heart weight; BW: body weight; LV: left ventricle; RV: right ventricle

Free Lung Cell and Macrophage Activation Markers (3m exposure)

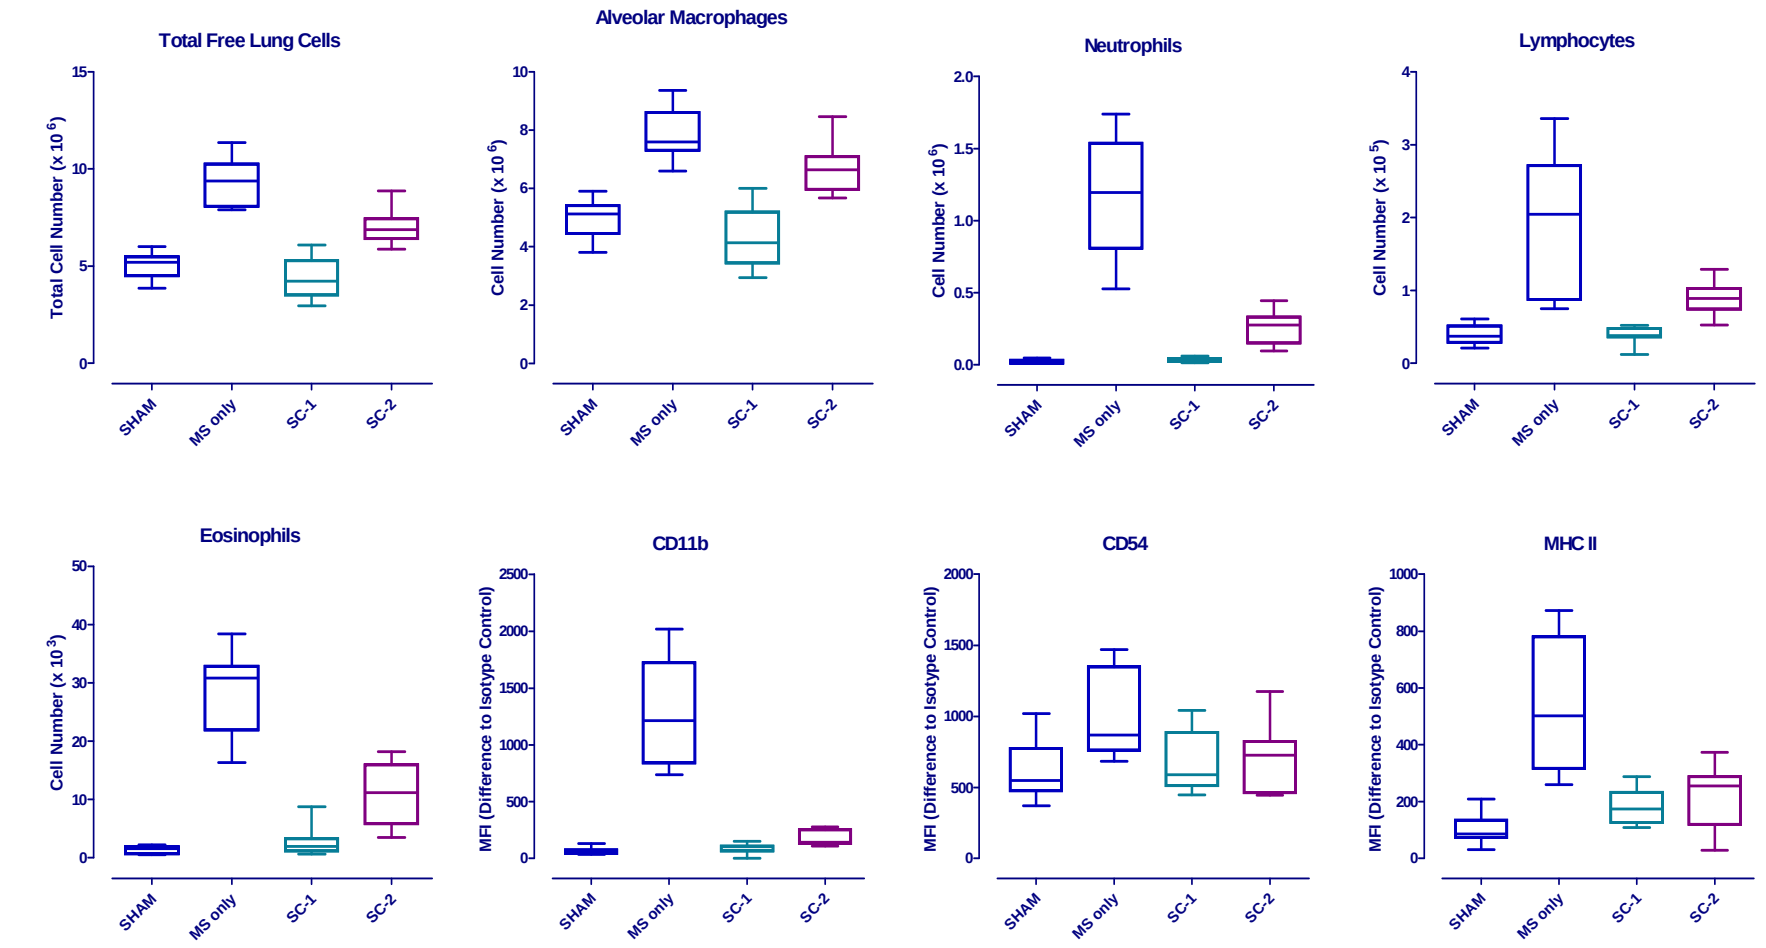
Total Free Lung Cells
Alveolar Macrophages
Neutrophils
Lymphocytes
Eosinophils
CD11b
CD54
MHC II
Total Cell Number (x 10^6)
Cell Number (x 10^6)
Cell Number (x 10^5)
Cell Number (x 10^3)
MFI (Difference to Isotype Control)
SHAM
MS only
SC-1
SC-2

MFI: mean fluorescence intensity

COPD-relevant Biomarkers in BALF (3m exposure)

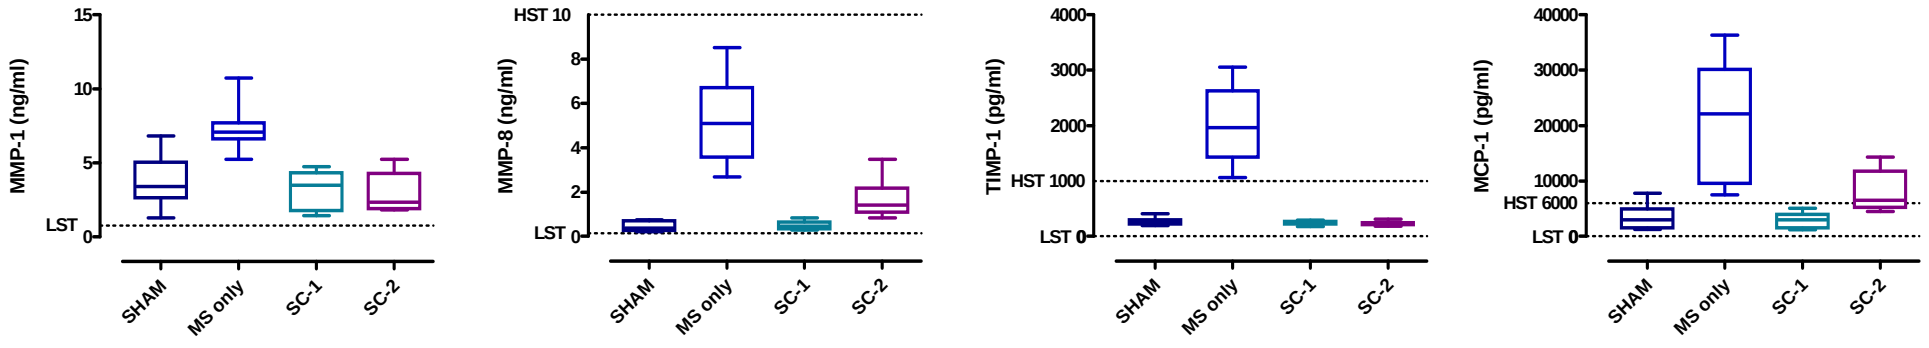
MMP-1 (ng/ml)
MMP-8 (ng/ml)
TIMP-1 (pg/ml)
MCP-1 (pg/ml)
LST
HST 10
HST 1000
HST 6000
SHAM
MS only
SC-1
SC-2

LST: lowest standard; HST: highest standard

Summary and Conclusion

- Cigarette smoke cessation resulted in a large increase in body weight.
 --Total concentration of nicotine metabolites in urine confirmed smoking cessation.
- Cigarette smoke increased cardiac weight (cardiac hypertrophy, normalization to tibia length).
 --Smoking cessation lowered the cardiac hypertrophy to the level of that of sham-exposed animals.
- Cigarette smoke increased the levels of MMP-1, MMP-8, TIMP-1, and MCP-1 in BALF.
 --Smoking cessation resulted in a complete (SC-1) or partial (SC-2) recovery of these biomarkers.
- Cigarette smoke induced pulmonary inflammation as detected by an increased influx of inflammatory cells (neutrophils, lymphocytes, alveolar macrophages, and eosinophils) into the alveolar space.
- Cigarette smoke induced an increased expression of the activation markers CD11b, CD54, and MHCII by alveolar macrophages.
 --Smoking cessation reversed most of the inflammatory changes completely (SC-1) or partially (SC-2).

Analysis of cardiovascular and pulmonary markers shows that the SH rat is a relevant model for cigarette-smoke-induced cardiac hypertrophy and lung inflammation. Hence, this experimental setup could be used to evaluate respiratory and cardiovascular effects of switching to potentially reduced-risk tobacco products after prior exposure to conventional cigarette smoke.

PMI RESEARCH & DEVELOPMENT

Society of Toxicology
50th Annual Meeting & ToxExpo™
Washington DC, USA
6-10 March 2011

Philip Morris International Research & Development, Quai Jeanrenaud 5, 2000 Neuchâtel, Switzerland
T: +41 58 242 21 11, **F**: +41 58 242 28 11, **W**: www.philipmorrisinternational.com