Genotoxicity of the gas/vapor phase (GVP) of cigarette mainstream smoke in the mouse lymphoma TK assay (MLA)

S. Wittke and E. Trelles-Sticken
Philip Morris International R&D, Philip Morris Research Laboratories GmbH, Cologne, Germany
SOT Sponsor: T. Mueller

Introduction and Objective

The mouse lymphoma TK assay (MLA, Clive et al., 1972), is a genotoxicity assay which measures the induction of forward mutations at the tk-locus in L5178Y/tk$^{+/-}$-3.7.2C mouse lymphoma cells. The MLA is generally used determine the mutagenicity of the particulate phase of cigarette smoke (total particulate matter [TPM]) (Schramke et al., 2006). To generate a comprehensive and thus more relevant image of cigarette-smoke-related mammalian mutagenicity *in vitro*, we extended this approach to the water-soluble portion of the gas/vapor phase (GVP).

The objective of this study was to validate the MLA for the characterization of GVP from conventional cigarettes.

Materials and Methods

Cigarettes and GVP collection

- University of Kentucky Reference Cigarette 3R4F (TPM yield ~12 mg/cig.).
- Cigarettes (12 per batch) were smoked on a 20-port Borgwaldt smoking machine in basic conformity with ISO standard 3308 (2000).
- TPM was filtered out by passing the whole smoke through glass fiber filters.
- The remaining GVP was bubbled through 36 ml ice-cold phosphate-buffered saline (PBS) in a gas wash bottle to trap the water-soluble portion.
- Reproducibility of smoke generation was checked by determining the amount of TPM and the amount of acrolein in GVP (maximum RSD: 3.6%)

Mutagenicity testing

- L5178Y/tk$^{+/-}$- 3.7.2C mouse lymphoma cells
- Microtiter plate version of the MLA (Cole et al., 1986) in basic accordance with OECD guideline 476 (1997)
- Exposure time: 4-h with and without exogenous metabolic promutagen activation (S9) and 24-h without S9 on separate days
- Expression times: 2 and 3 days
- Cytotoxicity measured by determination of the relative total growth (RTG) of the cultures after the treatment
- Seeding of cells
 --2000 cells per well to determine mutant frequency (MF) in selective medium containing TFT (4 µg/ml)
 --1 cell per well in none-selective medium without TFT to determine cloning efficiency (CE; viability)
- Colony detection after 10-14 days
 --incubation of the colonies in the wells with the fluorescence viability indicator resazurin (0.14 mg/ml) for 16 to 32 h at 37.0 ± 1 °C.
- Determination of fluorescence intensity in each well as a measure for proliferating colonies using a fluorescence microtiter plate reader
- MF was derived from the number of wells with mutant colonies in selective culture medium and the number of wells with colonies in nonselective culture medium

Statistical Analysis

- Dose-response curves were calculated by nonlinear regression analysis with with the power function $y = a + bx^c$
- Equal mutagenic effect concentration for 3 times the background (spontaneous) mutant frequency (C_{3B}) was calculated
 --the lower the C_{3B} value the higher the mutagenic activity
- Repeatability and reproducibility were determined for MF and $1/C_{3B}$ values according to ISO standard 5725-2 (1994, 2002)

Results

Response of 3R4F-GVP, 24-h Exposure, 2-d Expression Time

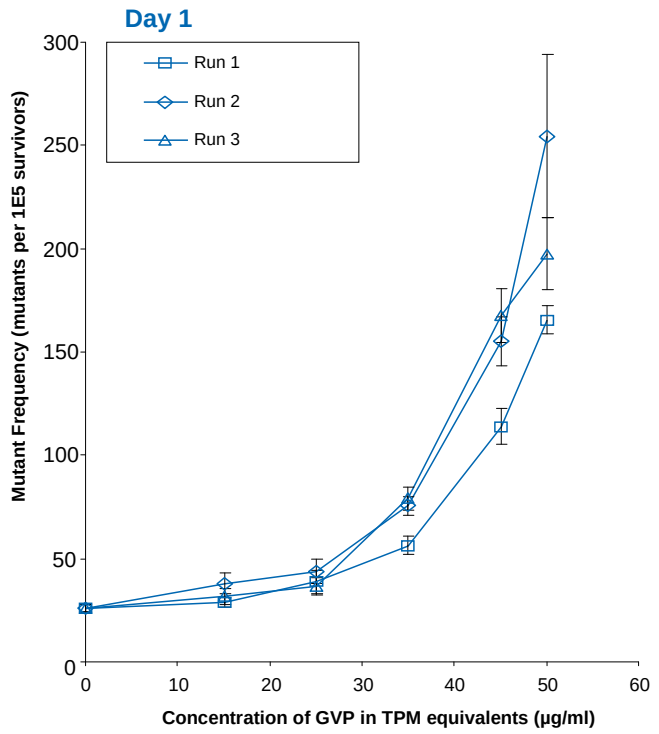
Day 1
Run 1
Run 2
Run 3
Mutant Frequency (mutants per 1E5 survivors)
Concentration of GVP in TPM equivalents (µg/ml)

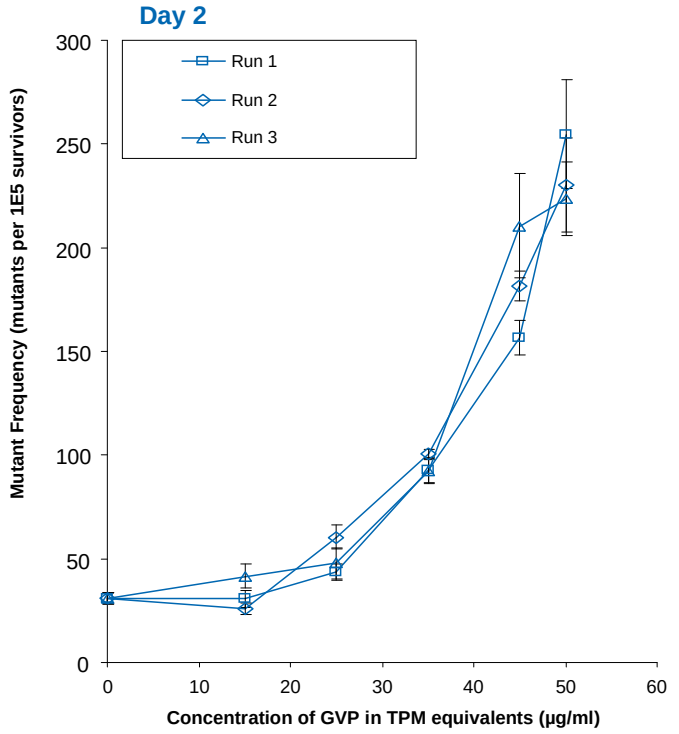
Day 2
Run 1
Run 2
Run 3
Mutant Frequency (mutants per 1E5 survivors)
Concentration of GVP in TPM equivalents (µg/ml)

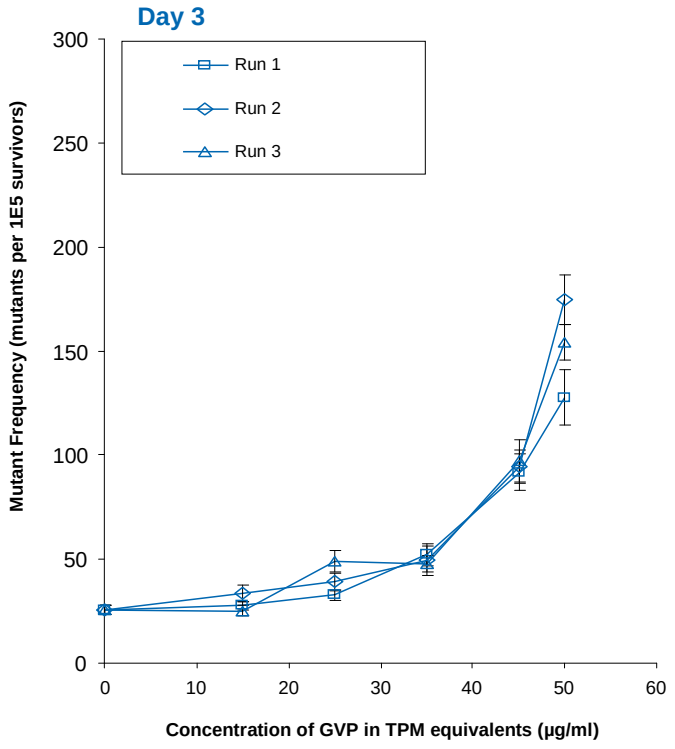
Day 3
Run 1
Run 2
Run 3
Mutant Frequency (mutants per 1E5 survivors)
Concentration of GVP in TPM equivalents (µg/ml)

Response of 3R4F-GVP, 4-h Exposure, 3-d Expression Time

Without S9

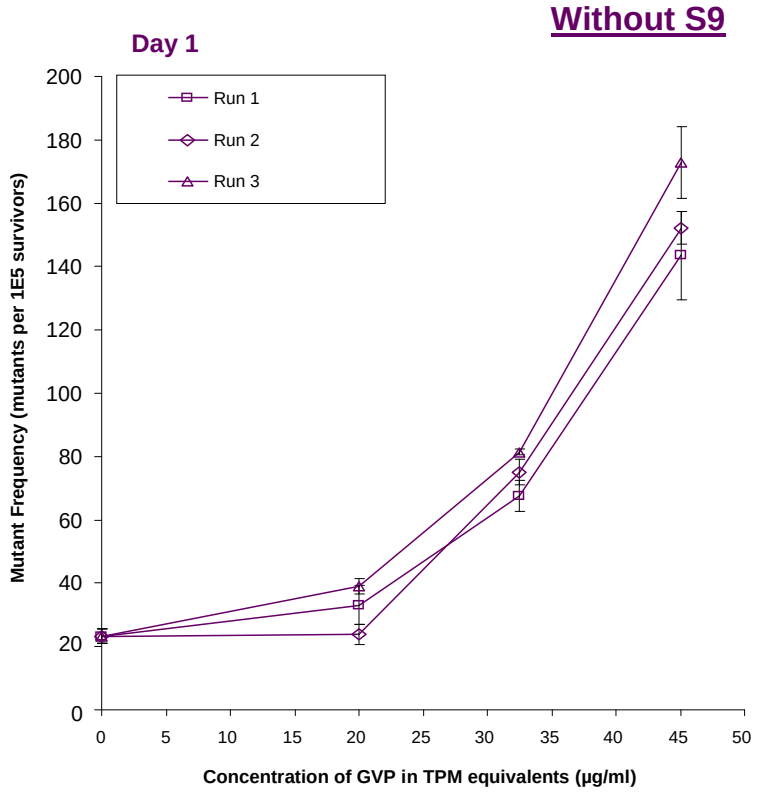
Day 1
Run 1
Run 2
Run 3
Mutant Frequency (mutants per 1E5 survivors)
Concentration of GVP in TPM equivalents (µg/ml)

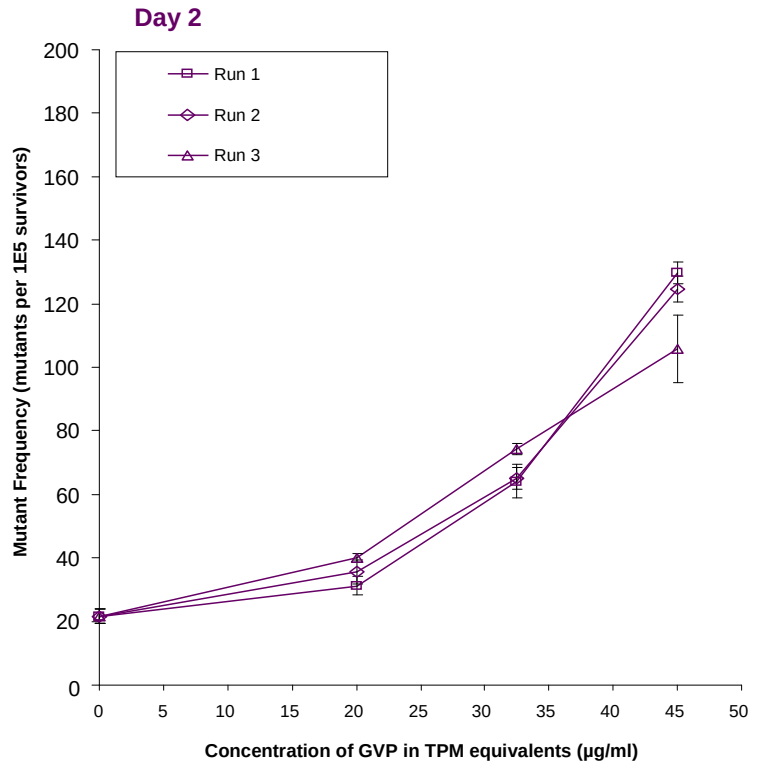
Day 2
Run 1
Run 2
Run 3
Mutant Frequency (mutants per 1E5 survivors)
Concentration of GVP in TPM equivalents (µg/ml)

With S9

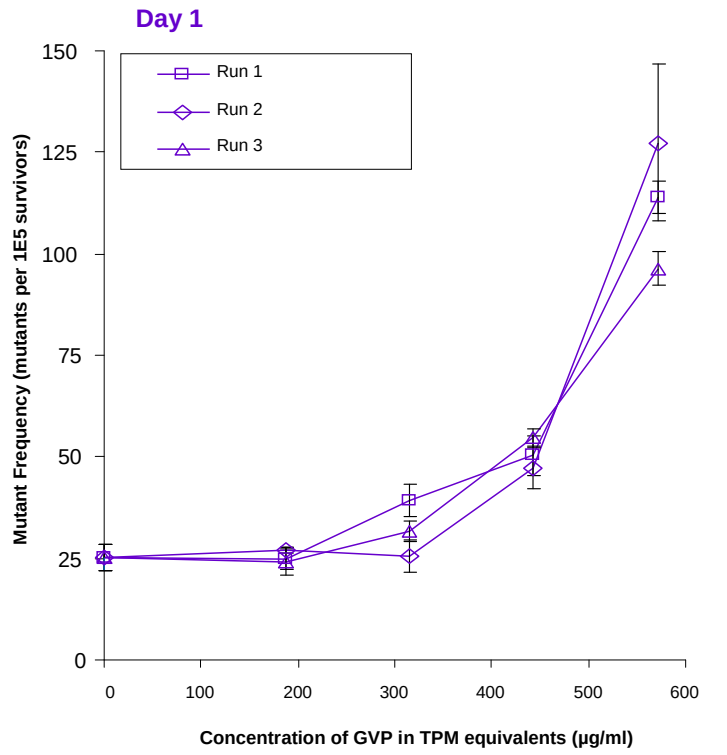
Day 1
Run 1
Run 2
Run 3
Mutant Frequency (mutants per 1E5 survivors)
Concentration of GVP in TPM equivalents (µg/ml)

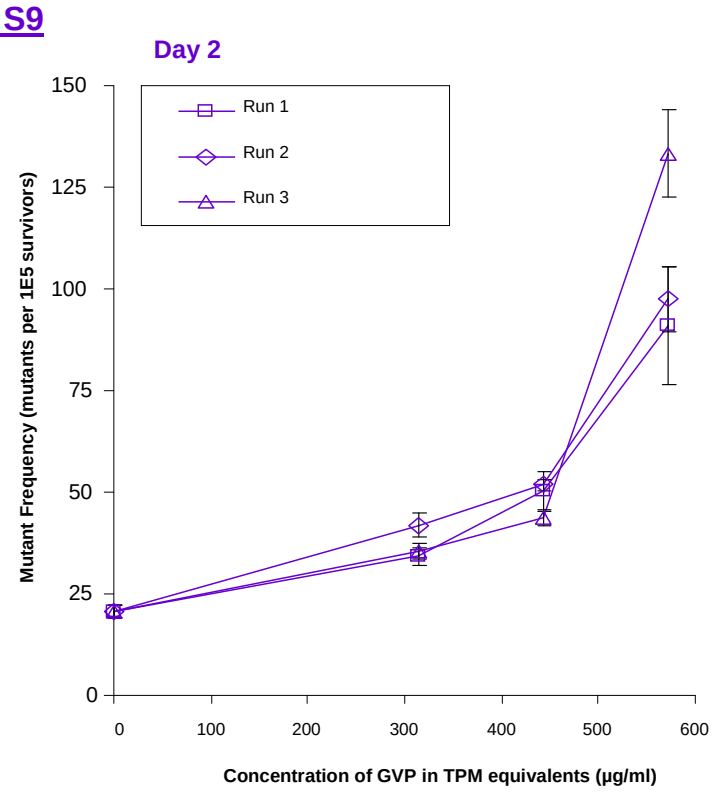
Day 2
Run 1
Run 2
Run 3
Mutant Frequency (mutants per 1E5 survivors)
Concentration of GVP in TPM equivalents (µg/ml)

Error bars represent mean ± SE from N = 5 plates

Variability

24-h Exposure, 2-d Expression Time

	Repeatability RSD (%)	Reproducibility RSD (%)
MF (mean)	15.5	26.1
$1/C_{3B}$ Value	5.8	7.8

4-h Exposure, 3-d Expression Time

		Repeatability RSD (%)	Reproducibility RSD (%)
MF (mean)	-S9	12.4	16.2
	+S9	11.9	14.2
$1/C_{3B}$ Value	-S9	4.9	4.9
	+S9	2.1	5.1

References
- Clive, D., et al., A mutational assay system using the thymidine kinase locus in mouse lymphoma cells. *Mutat Res*. 1972 Sep;16(7):77-87.
- Cole, J. et al., The mutagenicity of sodium fluoride to L5178Y [wild-type and TK$^{+/-}$ (3.7.2c)] mouse lymphoma cells. *Mutagenesis*. 1986 Mar;1(2):157-67.
- International Organization for Standardization (ISO):
 --ISO 3308, Routine analytical cigarette-smoking machine - Definitions and standard conditions, 4th ed., 2000.
 --ISO 5725-2, Accuracy (trueness and precision) of measurement methods and results – Part 2: A basic method for the determination of repeatability and reproducibility of a standard measurement method, 1994, 2002 (Technical Corrigendum).
- OECD: Guideline 476. In vitro mammalian cell gene mutation test, in: OECD Guidelines for Testing of Chemicals, Paris: Organization for Economic Co-operation and Development, 1997.
- Schramke et al., The mouse lymphoma thymidine kinase assay for the assessment and comparison of the mutagenic activity of cigarette mainstream smoke particulate phase. *Toxicology*. 2006 Oct 29;227(3):193-210.

Summary and Conclusion

- Clear-cut dose-response relationships were found for MF under all assay conditions.
- 24-h treatment with 2-d expression time and 4-h treatment (with and without S9) with 3-d expression time showed good repeatability and reproducibility in terms of mean MF values and $1/C_{3B}$ values.

The MLA is sensitive for the detection of the *in vitro* mammalian mutagenicity of the gas/vapor phase of cigarette mainstream smoke.

PMI RESEARCH & DEVELOPMENT

Society of Toxicology
50th Annual Meeting & ToxExpo™
Washington DC, USA
6-10 March 2011

Philip Morris International Research & Development, Quai Jeanrenaud 5, 2000 Neuchâtel, Switzerland
T: +41 58 242 21 11, **F**: +41 58 242 28 11, **W**: www.philipmorrisinternational.com