6-Aryl-β-Carbolines as Potent Tubulin-Polymerization Inhibitors

Paul Blaney[a], Simon Foster[a], Emma Blaney[a], Seb Hallworth[a], Andy Smith[a], Paul Doyle[a], Ray Fisher[a], Axel Teichert [b], Gerhard Lang[b], Stephane Demotz[b], Damien McHugh[b], Pavel Pospisil[b], Fernando Goffman[b]

[a] Peakdale Molecular Ltd., Chapel-en-le-Frith, Derbyshire, SK23 0PG
[b] Philip Morris Products S.A., Quai Jeanrenaud 5 CH-2000, Neuchatel, CH

1 SUMMARY

Natural products have proven to be an excellent resource for discovering new potential drug candidates. For example, a recent study has shown that 64% of anti-cancer drugs and 76% of anti-bacterial drugs are natural products or derivatives.[1]

Within a program aimed at the exploration of the biological activities of semisynthetic libraries based on natural product scaffolds – one of which was harmane (**1**) – 6-aryl-β-carbolines were identified as potent cytotoxic agents with an in-vivo anti-tumour activity. The underlying mechanism was found to be microtubuli depolymerization by interaction with the colchicine binding site.[2]

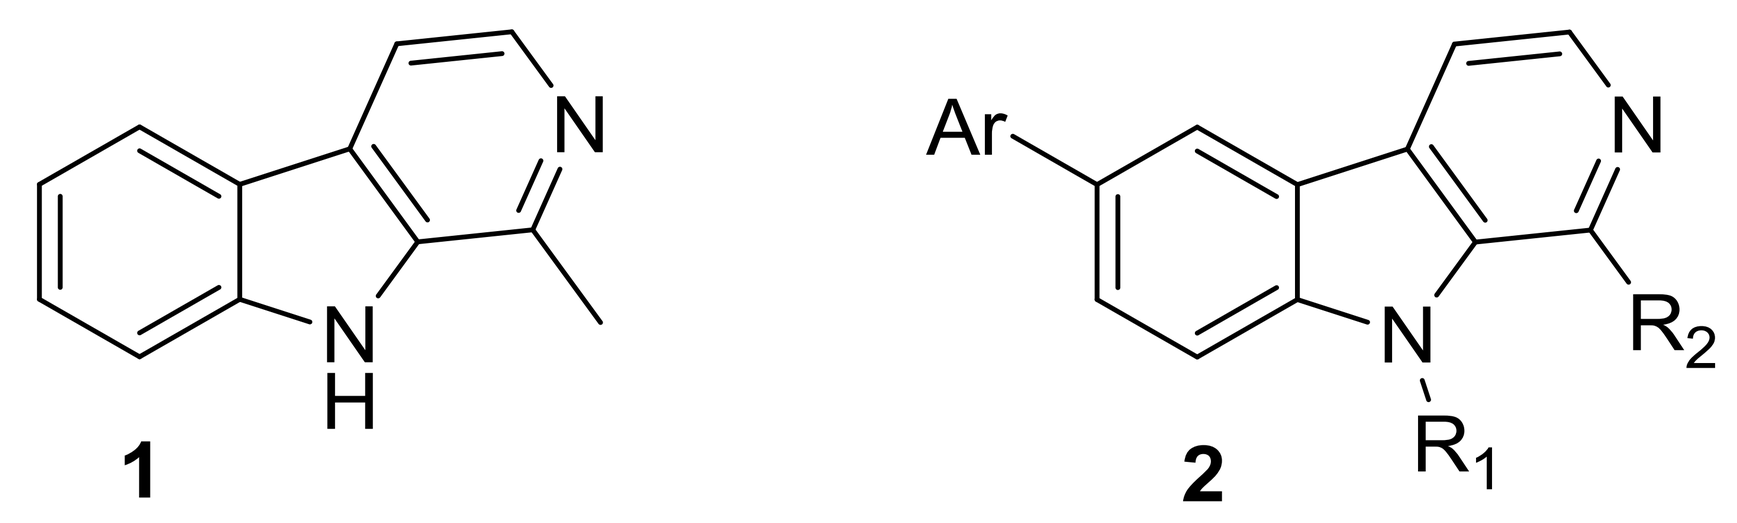
N
N
H
Ar
N
N
R2
R1

1 **2**

2 INTRODUCTION

Harmala β-carboline alkaloids were originally extracted from the plant *Peganum harmala*, commonly known as Syrian Rue, which has been used in herbal remedies in the Middle East, North Africa and China.

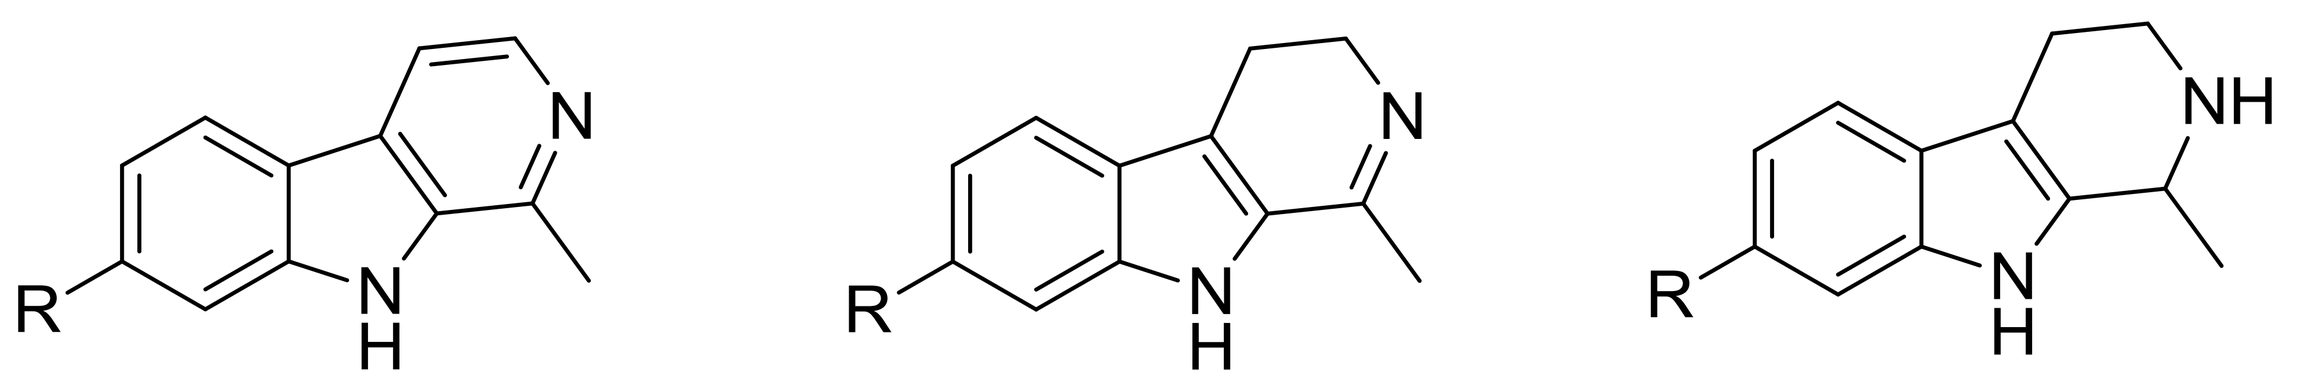

R = H, Harmane
R = OMe, Harmine
R = OH, Harmol

R = H, Harmalan
R = OMe, Harmaline
R = OH, Harmalol

R = OMe, Tetrahydroharmaline

Analogues of harmine were recently demonstrated to possess anti-tumour activity and β-carbolines were also shown to be highly cytotoxic, demonstrating apoptotic effect.[3] Further studies suggested β-carbolines could inhibit DNA topoisomerase I and II as well as CDK2 and CDK5, a cell regulation kinase.[4] Therefore, developing routes to analogues of harmane could give the potential for the discovery of novel compounds with anti-cancer activity.

3 SYNTHESIS OF HARMANE ANALOGUES – Initial Array Synthesis

The initial project involved array synthesis around several natural product templates, which included harmane. Over 1000 compounds were made around 7 different natural product templates in a 6 month period. Only the harmane work is presented here.

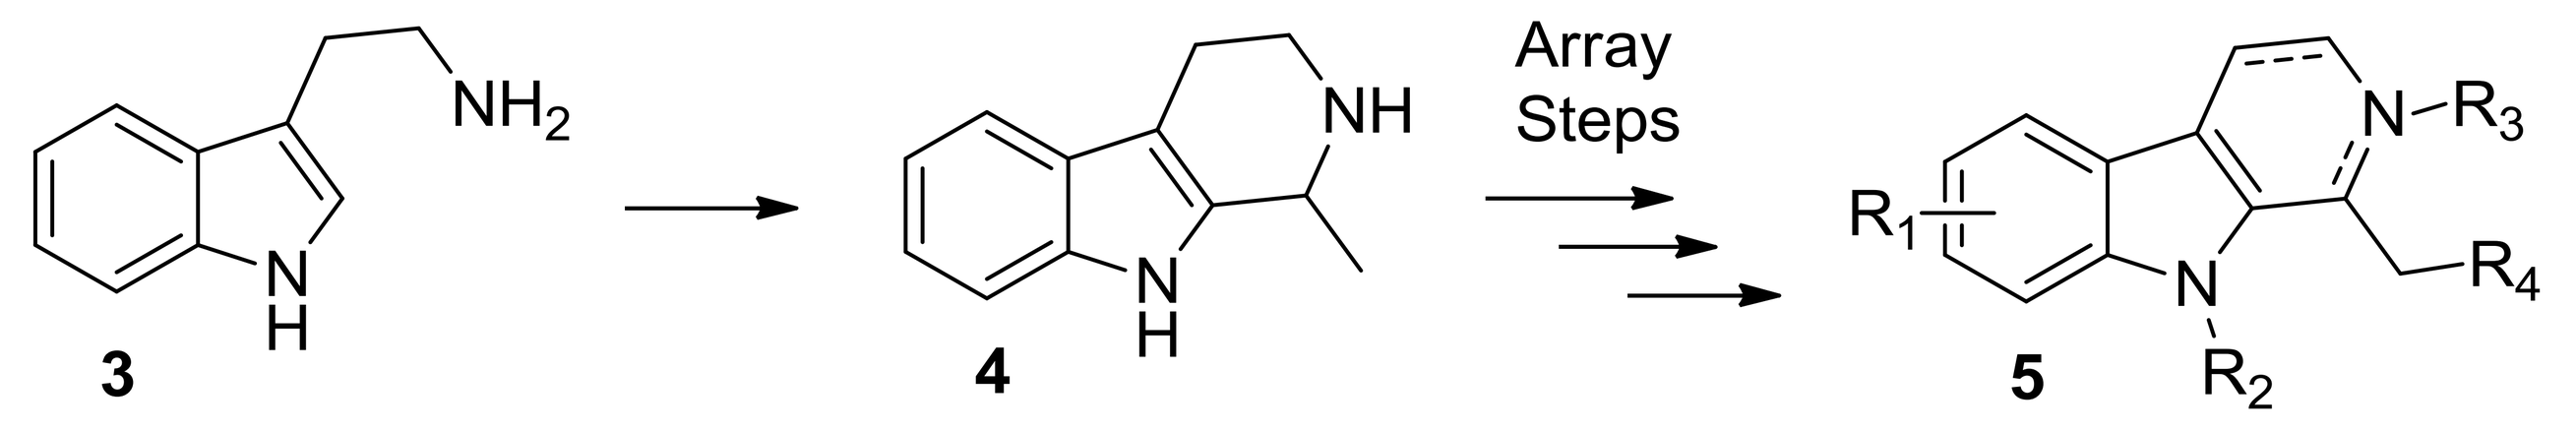
NH2
NH
Array
Steps
N-R3
R1
R4
R2
3
4
5

Starting from tryptamine **3**, a Pictet-Spengler cyclisation with acetaldehyde gave tetrahydroharmane **4**, which was then functionalised in a large variety of ways to give an array of over 300 compounds with the general structure shown as **5**. All compounds made were novel. A diversity analysis was conducted by Peakdale and the compounds were clustered using BCUT descriptors for screening.

A cytotoxicity screen on Jurkat cells identified three 6-aryl-β-carbolines with nanomolar IC_{50}'s:

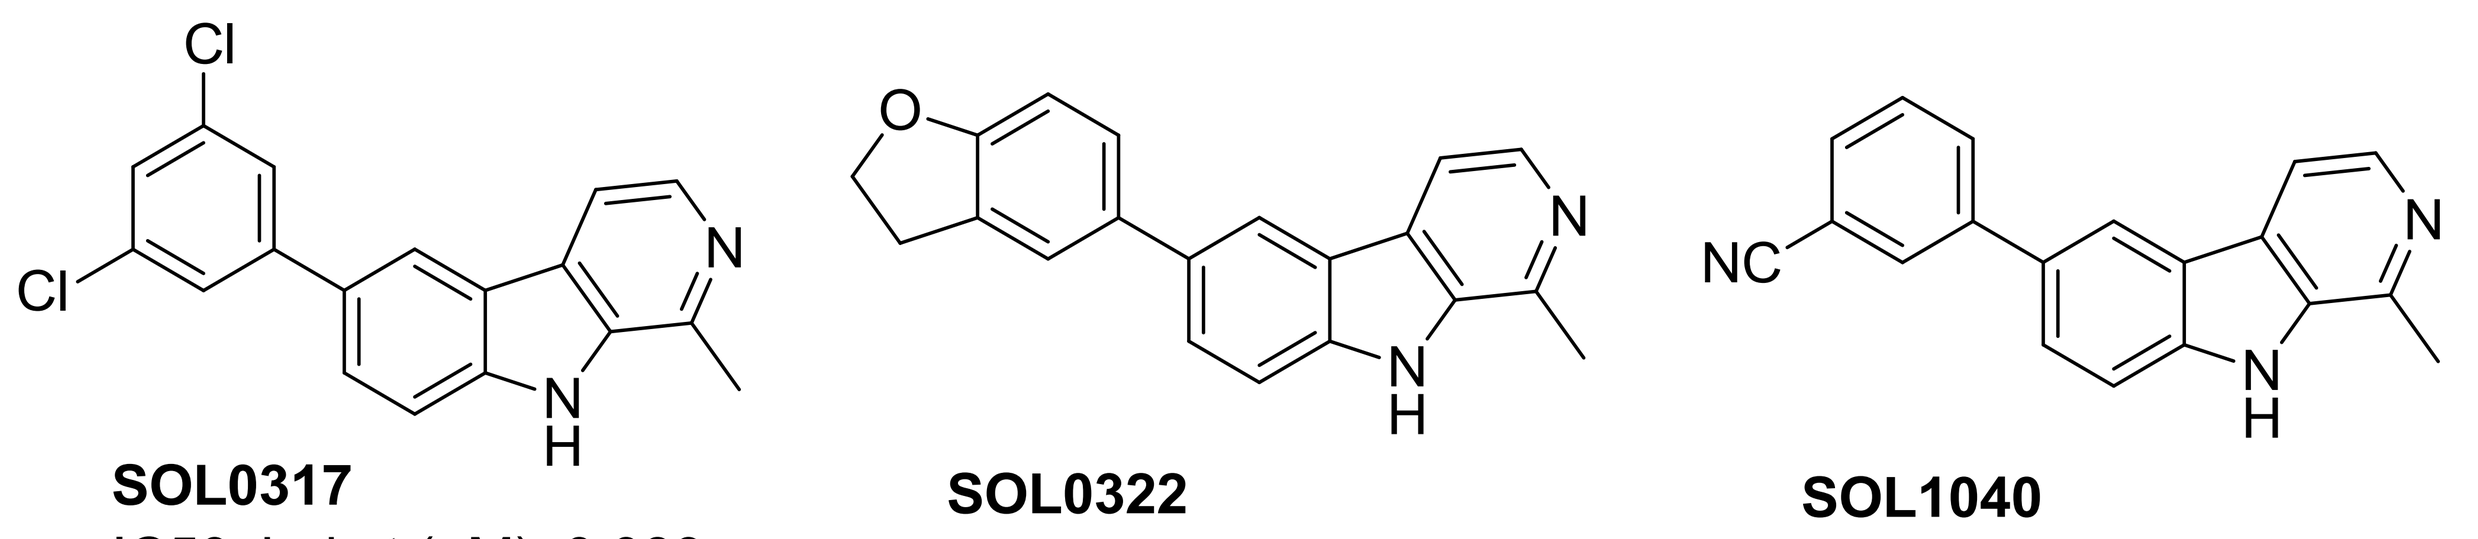
Cl
Cl
NC

SOL0317
IC50 Jurkat (μM): 0.089

SOL0322
IC50 Jurkat (μM): 0.064

SOL1040
IC50 Jurkat (μM): 0.047

A screen against a panel of kinases showed weak activity (~1 μM against Ser/Thr-kinases) only for **SOL0317**, while other cytotoxic analogues were inactive. No activity was found against DNA topoisomerases, which are known to be inhibited by some harmine analogues.

Active and inactive derivatives were then tested in caspase assays and the IC_{50} results obtained were largely overlapping with cytotoxicity results, suggesting that cytotoxicity was likely mediated by apoptosis. A number of further studies indicated that the compounds inhibited tubulin polymerisation (Figure 2) with competitive binding at the colchicine binding site.

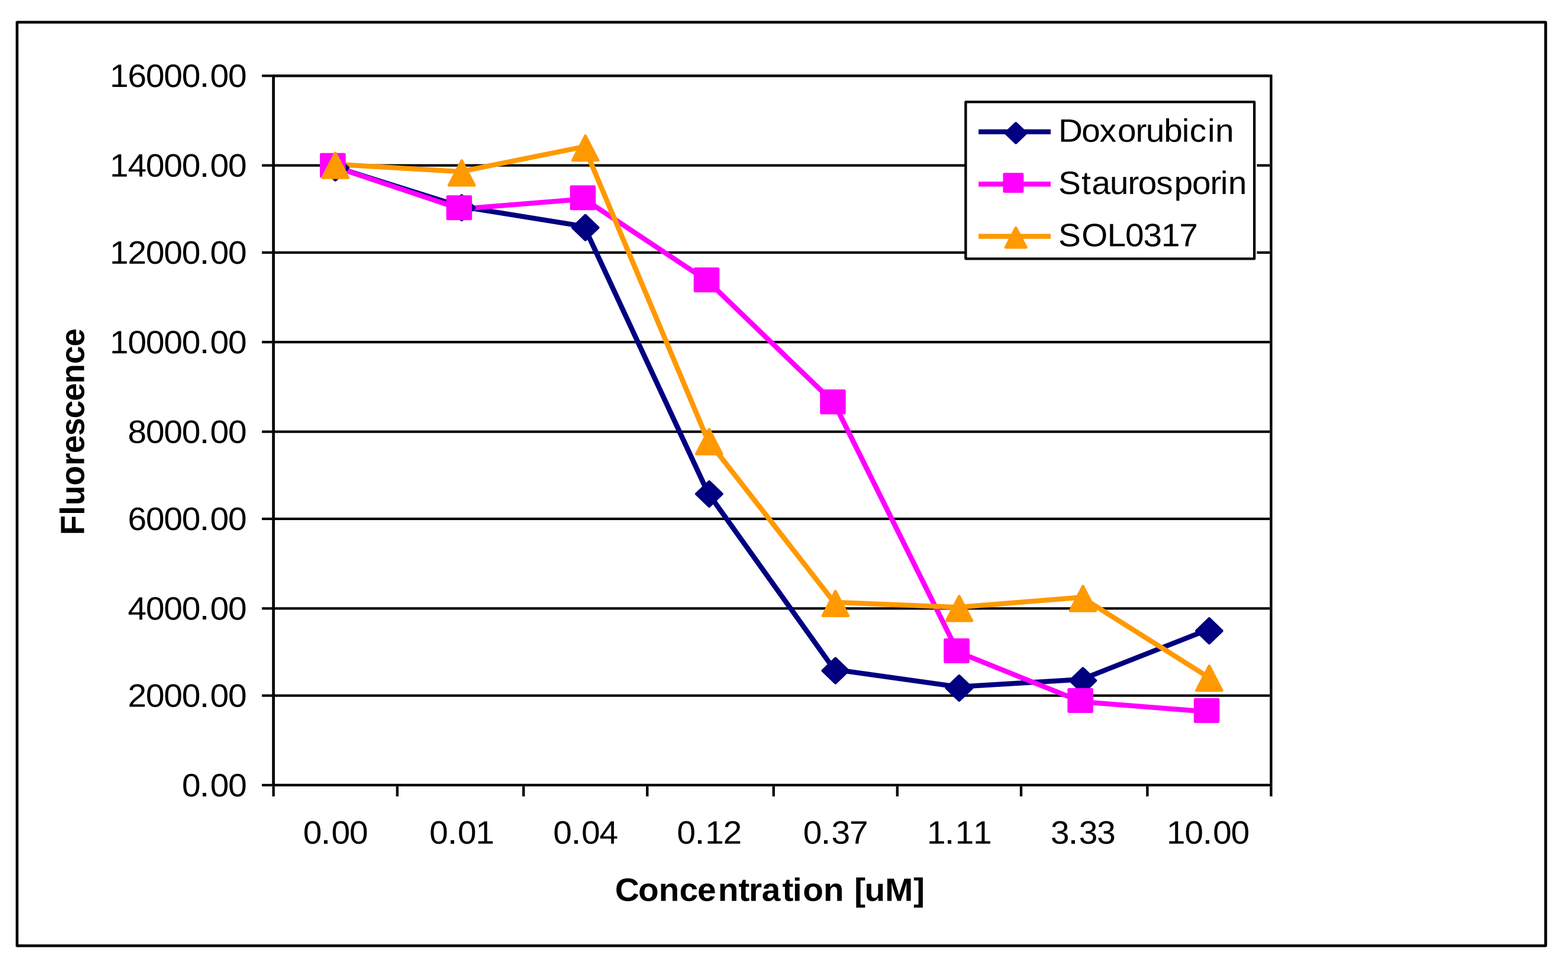
16000.00
14000.00
12000.00
10000.00
8000.00
6000.00
4000.00
2000.00
0.00
Fluorescence
0.00 0.01 0.04 0.12 0.37 1.11 3.33 10.00
Concentration [uM]
Doxorubicin
Staurosporin
SOL0317

Figure 1

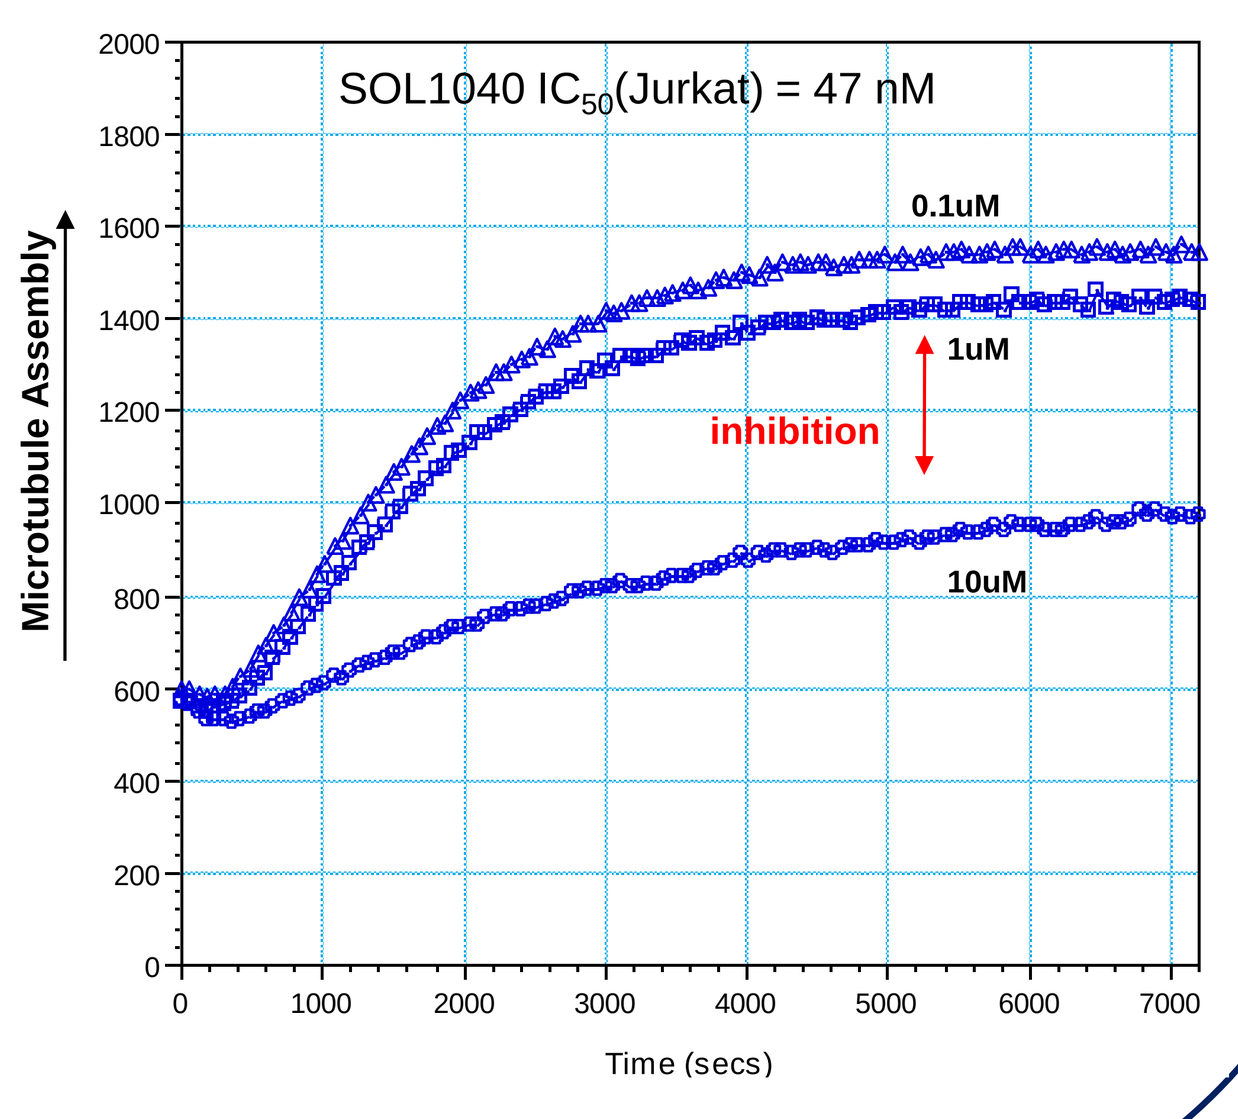
SOL1040 IC50(Jurkat) = 47 nM
0.1uM
1uM
inhibition
10uM
Microtubule Assembly
Time (secs)
0 1000 2000 3000 4000 5000 6000 7000
2000 1800 1600 1400 1200 1000 800 600 400 200 0

Figure 2

4 HARMANE ANALOGUES – Lead Optimisation

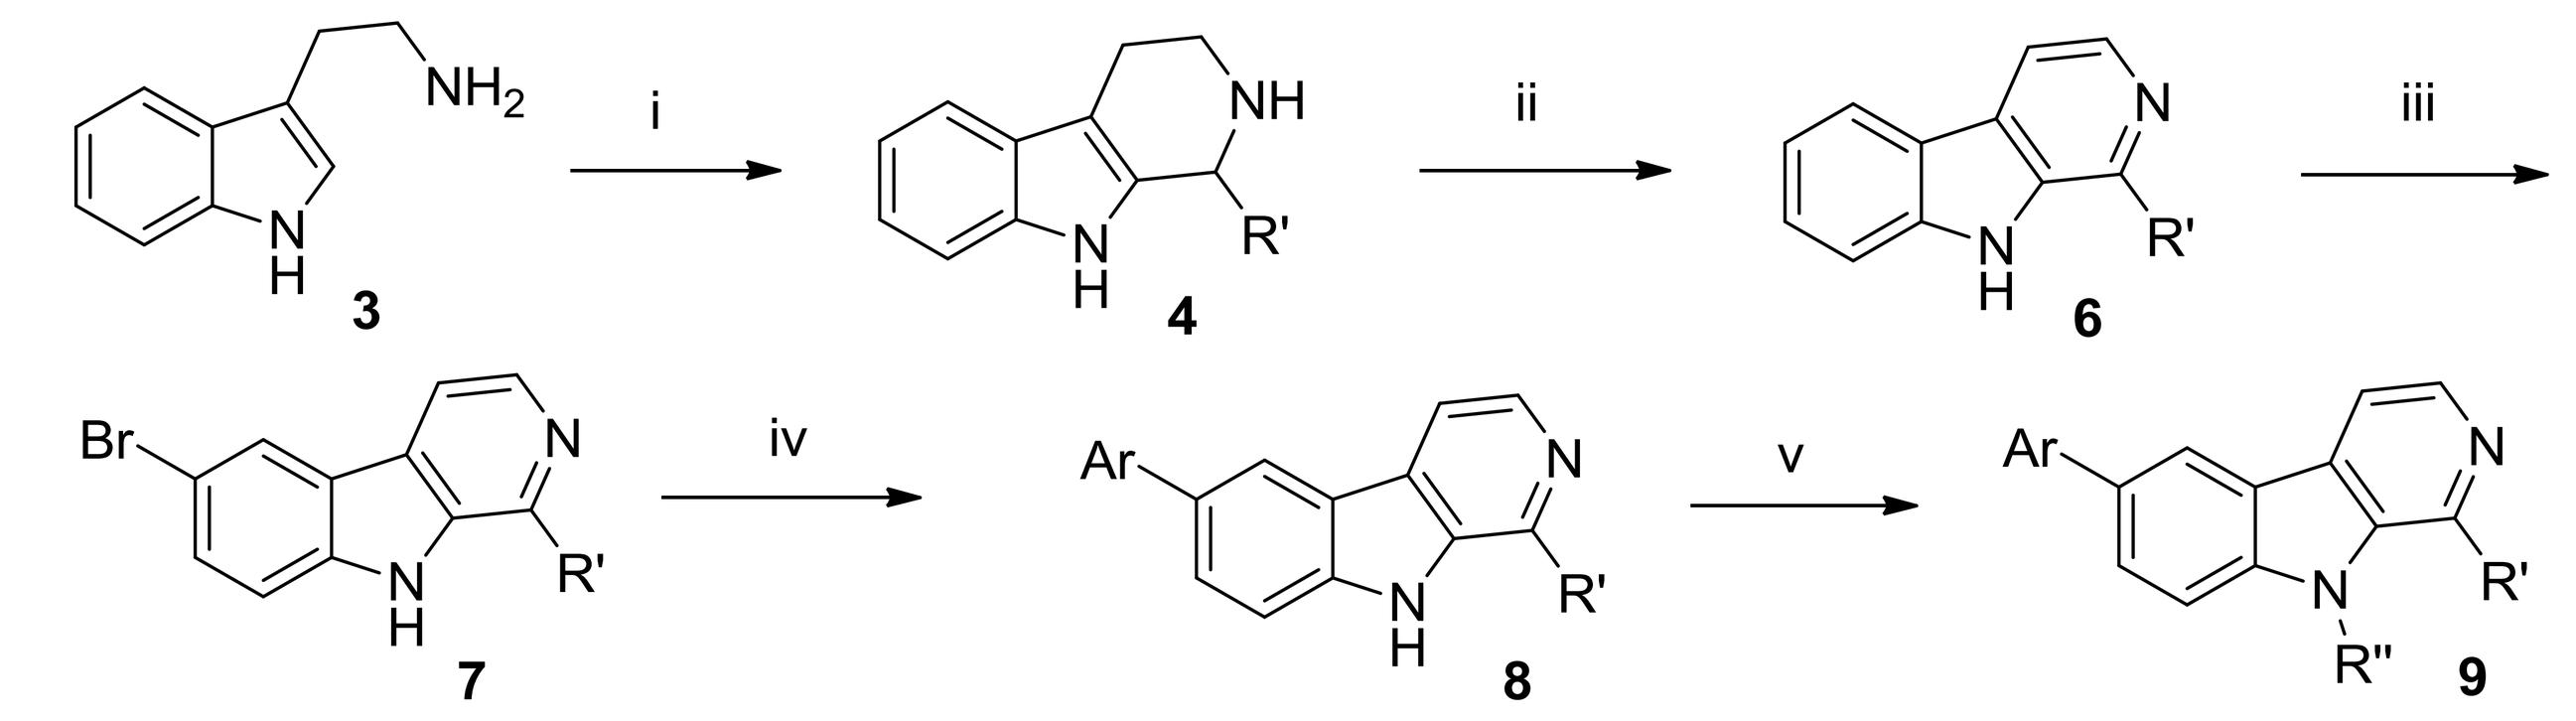
NH2
i
NH
R'
ii
iii
3
4
6
Br
iv
Ar
v
R''
7
8
9

i. R'CHO, dil. H_2SO_4, reflux, 55-70%; **ii.** 5% Pd on C, Xylene, 140ºC, 45%; **iii.** NBS, DCM, RT, 31%;
iv. $ArB(OR)_2$, $PdCl_2dppf$, K_3PO_4, 1,4-Dioxane, H_2O, 90ºC, 24-90%; **v.** NaH, R''Br or R''I, DMF, RT, 89%

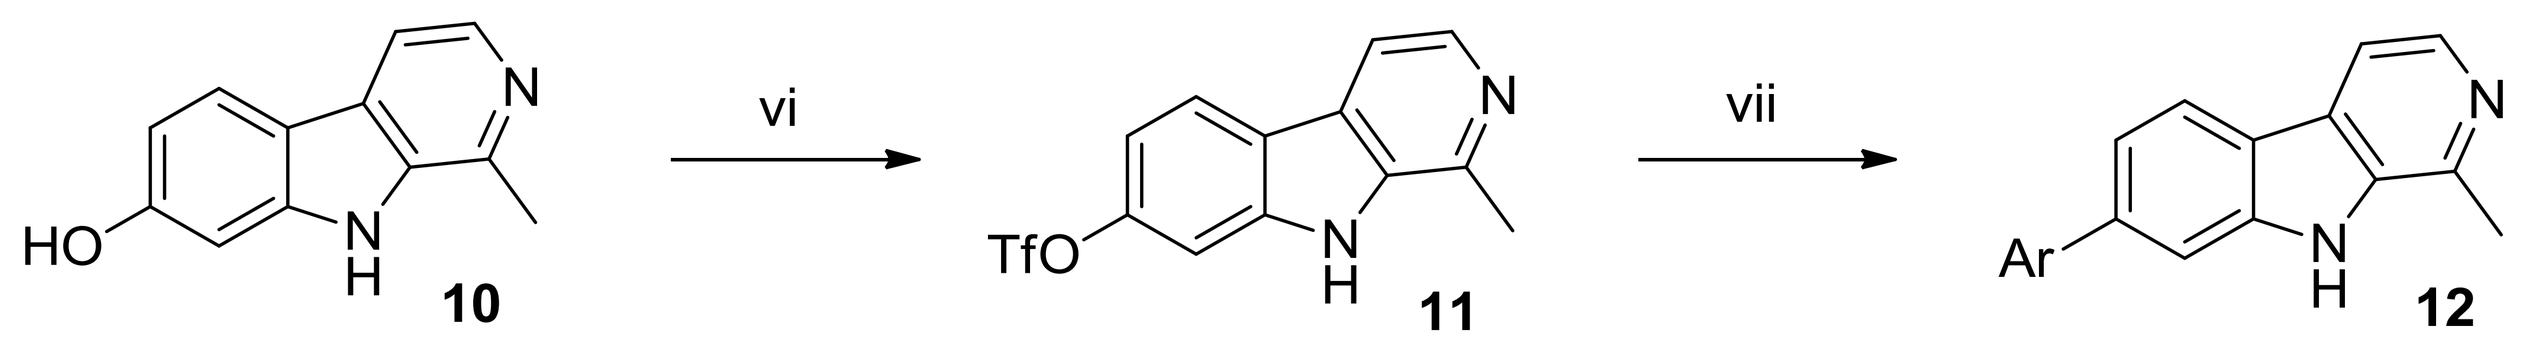
HO
vi
TfO
vii
Ar
10
11
12

vi. $PhN(OTf)_2$, Et_3N, DMF, RT, 79%; **vii.** $ArB(OR)_2$, $PdCl_2dppf$, K_3PO_4, 1,4-Dioxane, H_2O, 90ºC, 34-51%

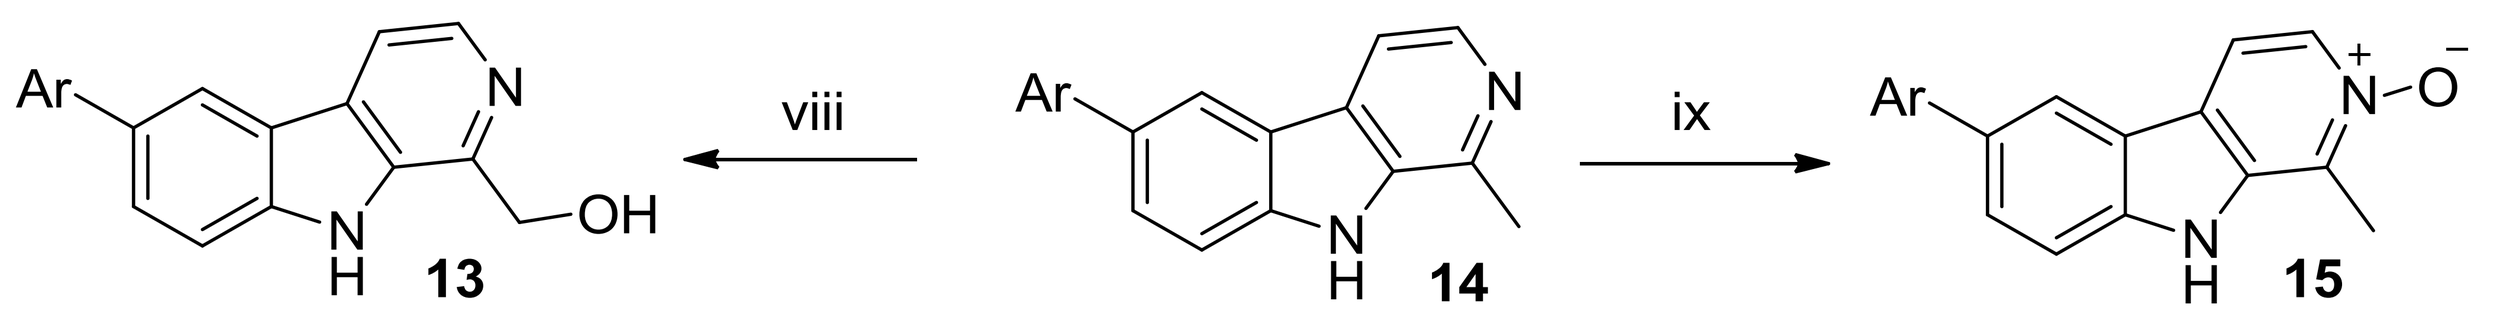
Ar
OH
viii
ix
13
14
15

viii. Ac_2O, MeCN, reflux, 84% then 2M NaOH, MeOH, reflux, 58%; **ix.** MCPBA, $CHCl_3$, RT, 50%

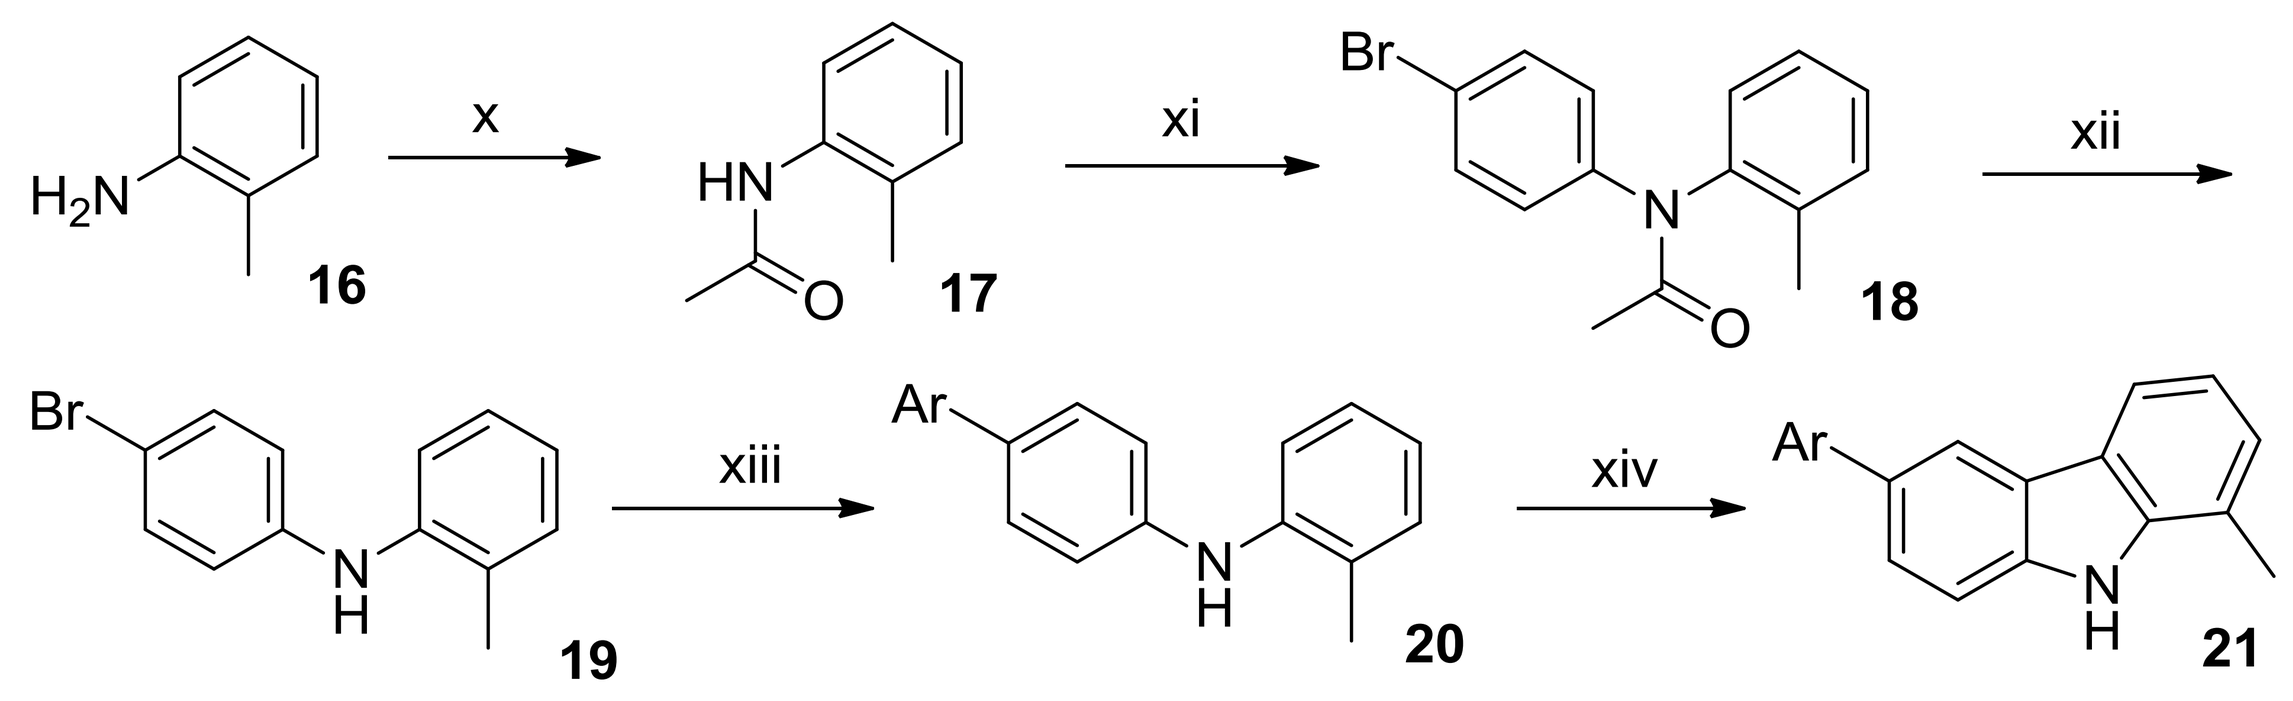
H2N
x
HN
xi
Br
xii
16
17
18
xiii
Ar
xiv
19
20
21

x. AcCl, pyridine, RT, 86%; **xi.** 1,4-diBr-benzene, Cu, I_2, NMP, 180ºC, 33%; **xii.** NaOMe, Toluene, 100ºC, 100%;
xiii. $ArB(OR)_2$, $PdCl_2dppf$, K_3PO_4, 1,4-Dioxane, H_2O, 90ºC, 27%; **xiv.** $Pd(OAc)_2$, Cs_2CO_3, toluene, AcOH, 100ºC, 19%

5 CYTOTOXICITY SAR ANALYSIS

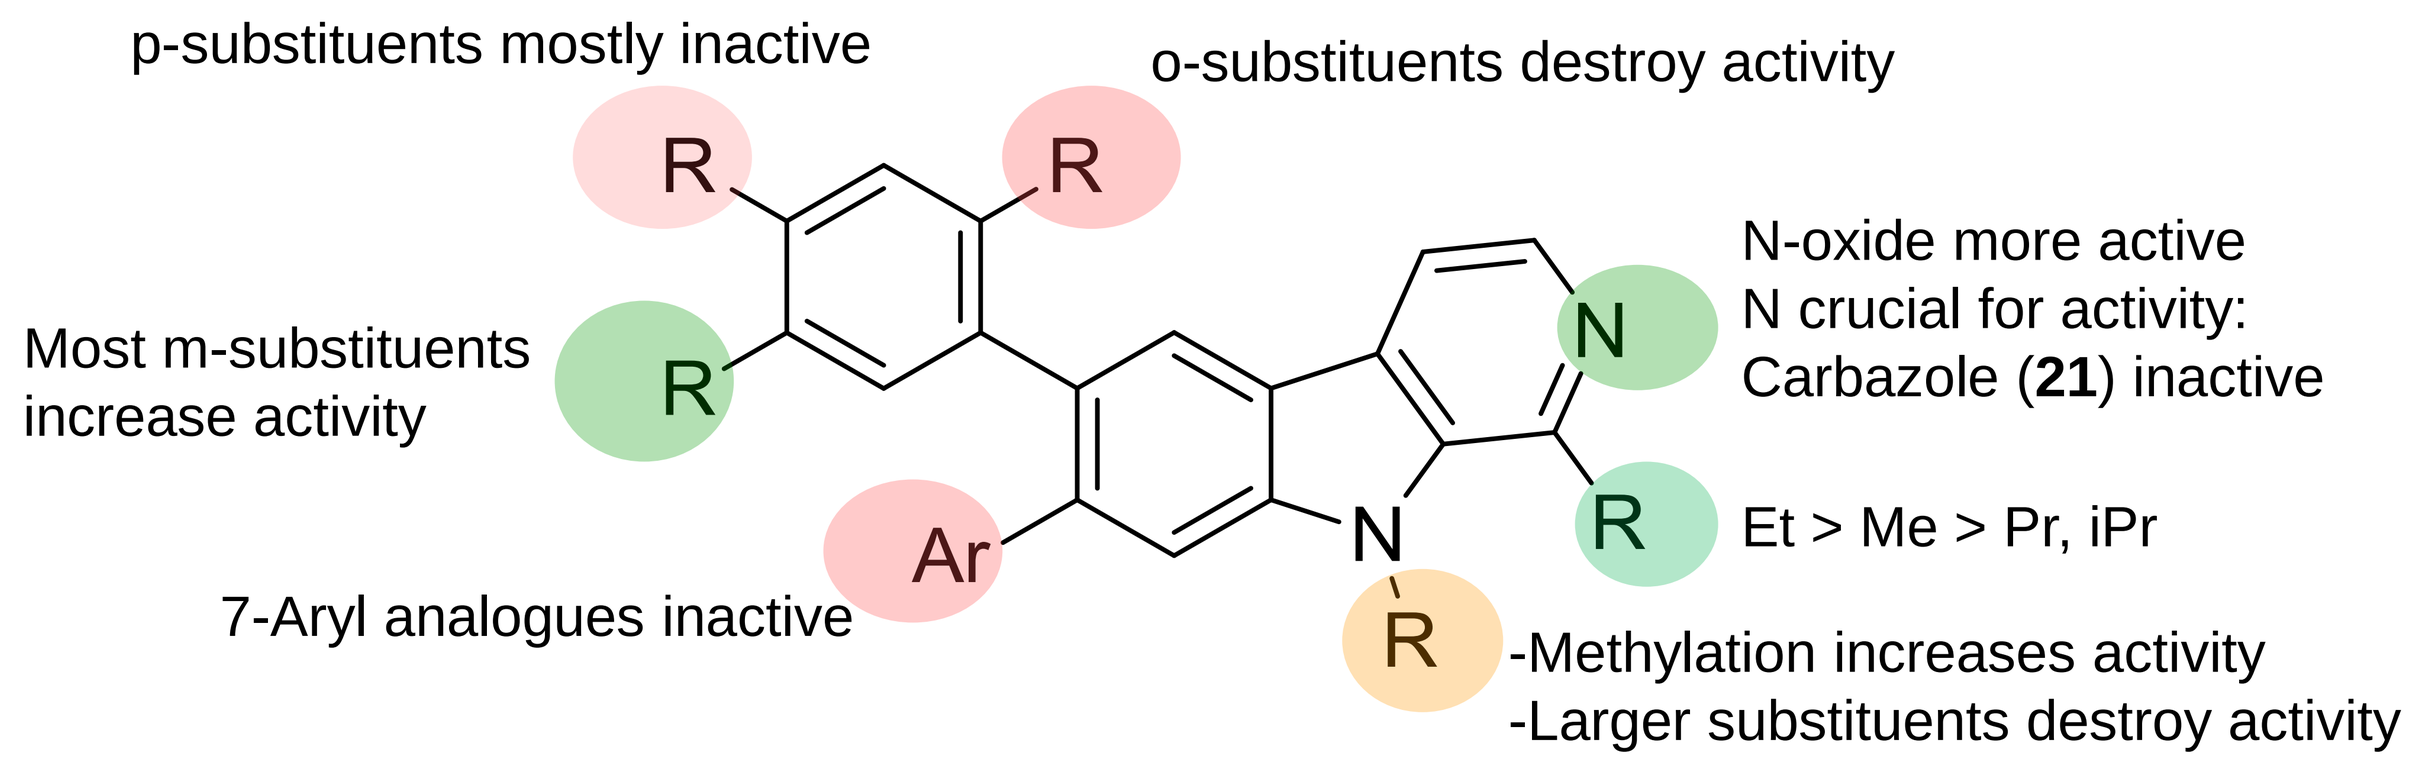
p-substituents mostly inactive
o-substituents destroy activity
Most m-substituents increase activity
N-oxide more active
N crucial for activity:
Carbazole (21) inactive
Et > Me > Pr, iPr
7-Aryl analogues inactive
-Methylation increases activity
-Larger substituents destroy activity

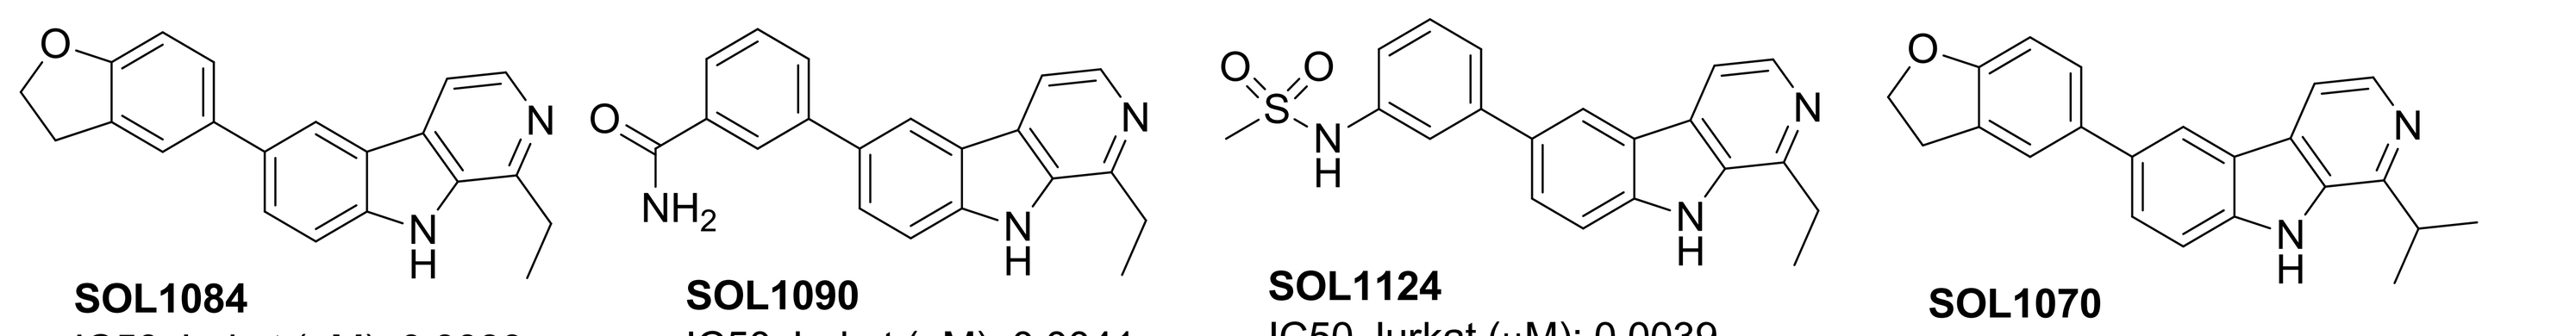

SOL1084
IC50 Jurkat (μM): 0.0039

SOL1090
IC50 Jurkat (μM): 0.0041

SOL1124
IC50 Jurkat (μM): 0.0039

SOL1070
IC50 Jurkat (μM): 0.3023

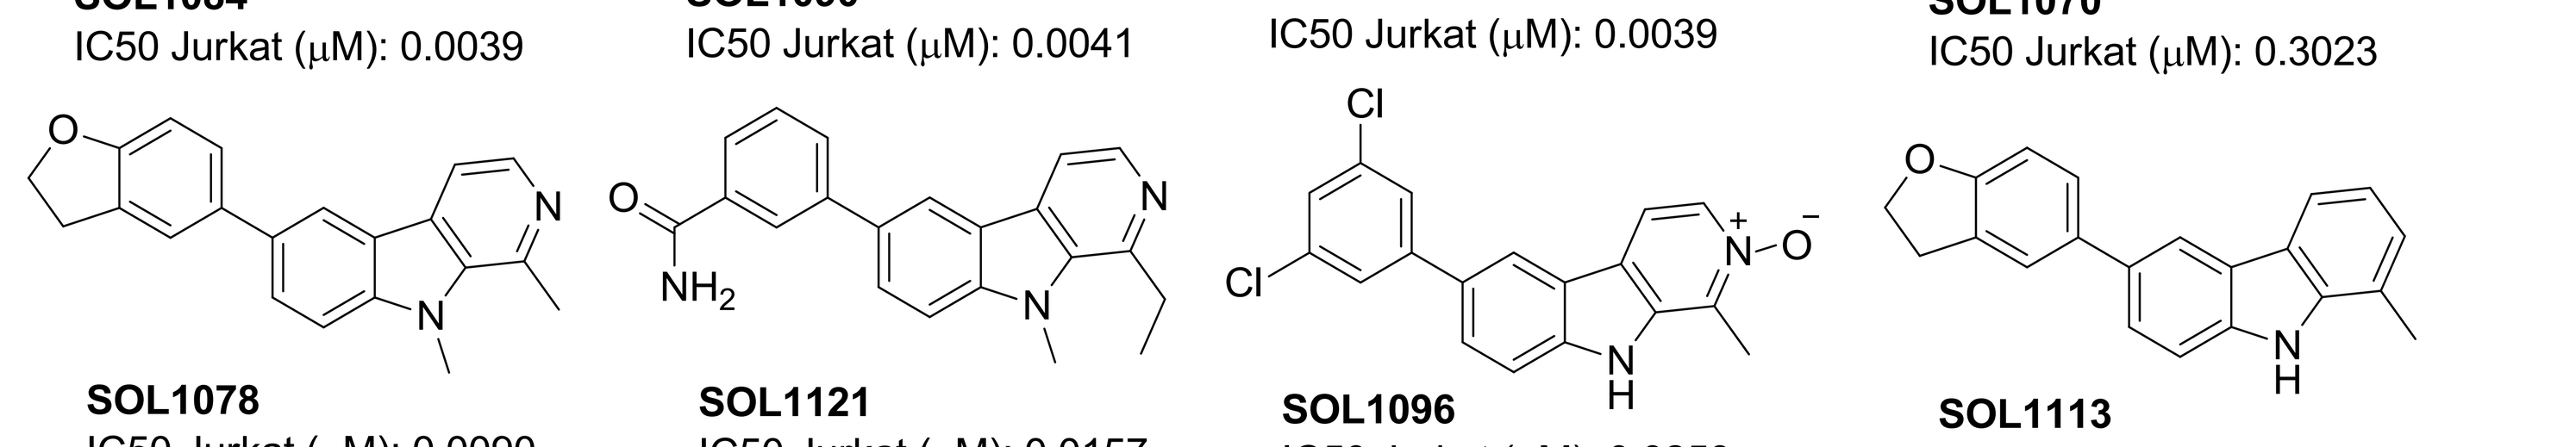

SOL1078
IC50 Jurkat (μM): 0.0090

SOL1121
IC50 Jurkat (μM): 0.0157

SOL1096
IC50 Jurkat (μM): 0.0253

SOL1113
IC50 Jurkat (μM): 3.7467

6 CONCLUSION

The β-carboline analogues presented in this poster have demonstrated activity against cancer cell lines. The compounds inhibit tubulin polymerisation by competitive binding at the colchicine binding site of the tubulin dimer. A number of the active analogues were tested against various tumour cell lines, of which hematopoietic cells were particularly sensitive. An increased time-to-endpoint (subject death or defined tumour volume) in mouse xenograft models for melanoma, colon carcinoma, fibrosarcoma and acute myelocytic leukaemia was observed upon treatment with selected compounds.

As the compounds act at the colchicine binding site on tubulin, they could be used in combination with the Vinca alkaloids and taxanes, which have two separate binding sites and could effect microtubule growth via a different mechanism. This project also demonstrates how natural products can be exploited as a starting point for drug discovery.

References

1. *J. Nat. Prod*., **2007**, *70*, 461 – 477.

2**. WO2012/059232**, Carbazole and Carboline Derivatives and Preparation and Therapeutic Applications Thereof.

3. *Int. J. Cancer*., **2004**, *114*, 675 – 682; *Current Med. Chem*., **2007**, *14*, 479 – 500.

4. *Pharmacol. Toxicol*., **2001**, *89*, 287 – 294; *Bioorg. Med. Chem. Lett*., **2001**, *11*, 1251 – 1255; *ibid*, **2002**, *12*, 1129 – 1132.

Acknowledgements

The authors wish to thank Philip Morris Products S.A. for their financial support.

Peakdale Molecular Ltd. Peakdale Science Park, Sheffield Road, Chapel-en-le-Frith, High Peak, SK23 0PG, UK | T +44(0)1298 816700 | E enquiries@peakdale.co.uk

peakdale.co.uk