OPTIMIZATION OF HISTOLOGICAL PROCESSING OF MURINE LUNG TISSUE

E. Seow[a], B. Phillips[a], E. Veljkovic[b], P. Vanscheeuwijck[b]

a Philip Morris International Research Laboratories, Pte. Ltd., 50 Science Park Road, Singapore 117406
b Philip Morris Products SA, Quai Jeanrenaud 5, CH-2000 Neuchâtel, Switzerland.

Introduction

This study has been conducted in order to understand the underlying cause of the lung tissue shrinkage and the alveolar collapse that has been observed in one of our pevious *in vivo* studies with C57BL/6 mice. We have evaluated the impact of the duration of storage in 70% ethanol and in 4% formaldehyde prior to the embedding in paraffin on the quality of the lung sections and the application of deliberate physical compression (a routine embedding procedure for some organs) of lungs samples.

Method

A total of 29 female 6-8 weeks old C57BL/6 mice bred under specific pathogen-free conditions were used for the study[1].
At necropsy whole lungs were fixed by intratracheal instillation with primary fixative ethanol-glycerol-acetic acid-formol-saline (EGAFS) for 48 hours. The duration of storage in 70% ethanol or 4% formaldehyde and the number of lungs used are summarized below.

Parameter	Duration in Weeks				
	1	3	6	9	12
70% Ethanol	4 left lungs	4 left lungs	4 left lungs	4 left lungs	4 left lungs
4% Formaldehyde	2 right lung lobes	2 right lung lobes	2 right lung lobes	2 right lung lobes	2 right lung lobes
Embedding-Compression	NA	NA	NA	NA	3 left lungs
Embedding-Without compression	NA	NA	NA	NA	6 left lungs

To evaluate the impact of the mechanical pressure, left lungs that were instilled with EGAFS for 48 hours and stored in 70% ethanol for 12 weeks were used; 6 left lungs were embedded in paraffin block without and 3 with deliberate pressure applied by using a “stamper”.

Both left and right lung lobes were processed and embedded in paraffin wax according to standard protocols[3]. Step-serial sections (4-µm thin) were then made from the paraffin-embedded right and left lung lobes at intervals of 150 µm, stained with haematoxylin-eosin and evaluated by the pathologist. The quality of the sections was evaluated based on the following parameters:

- The presence of the artificial folding of the pleura (irregularity of the lung surface);
- The presence of thickened and dense alveolar interstitium (localized centrally or multifocal);
- Number of step- serial sections made at a distance of 150µm;
- Dimension of the lung section displaying the main bronchus

Results

Several observations could be drawn from the above study:

- Tissue alterations including shrinkage were noted in almost all lung sections examined, independent of the duration of storage time and the solution used (Fig. 1-6);
- The correlation between the duration of the storage and the level of the alterations observed could not be established;
- Dimension of the left lung section displaying the main bronchus was similar among the lungs independent of the fixation period (Fig. 7)
- The number of step serial sections (9-17 levels) produced from each lung (Fig. 8) was generally lower than lungs with normal morphology (18-24 levels);
- In all sections generated from deliberate compressed lungs, tissue alteration (shrinkage) was observed (Fig. 9) compared to lung tissue without compression (Fig. 10)

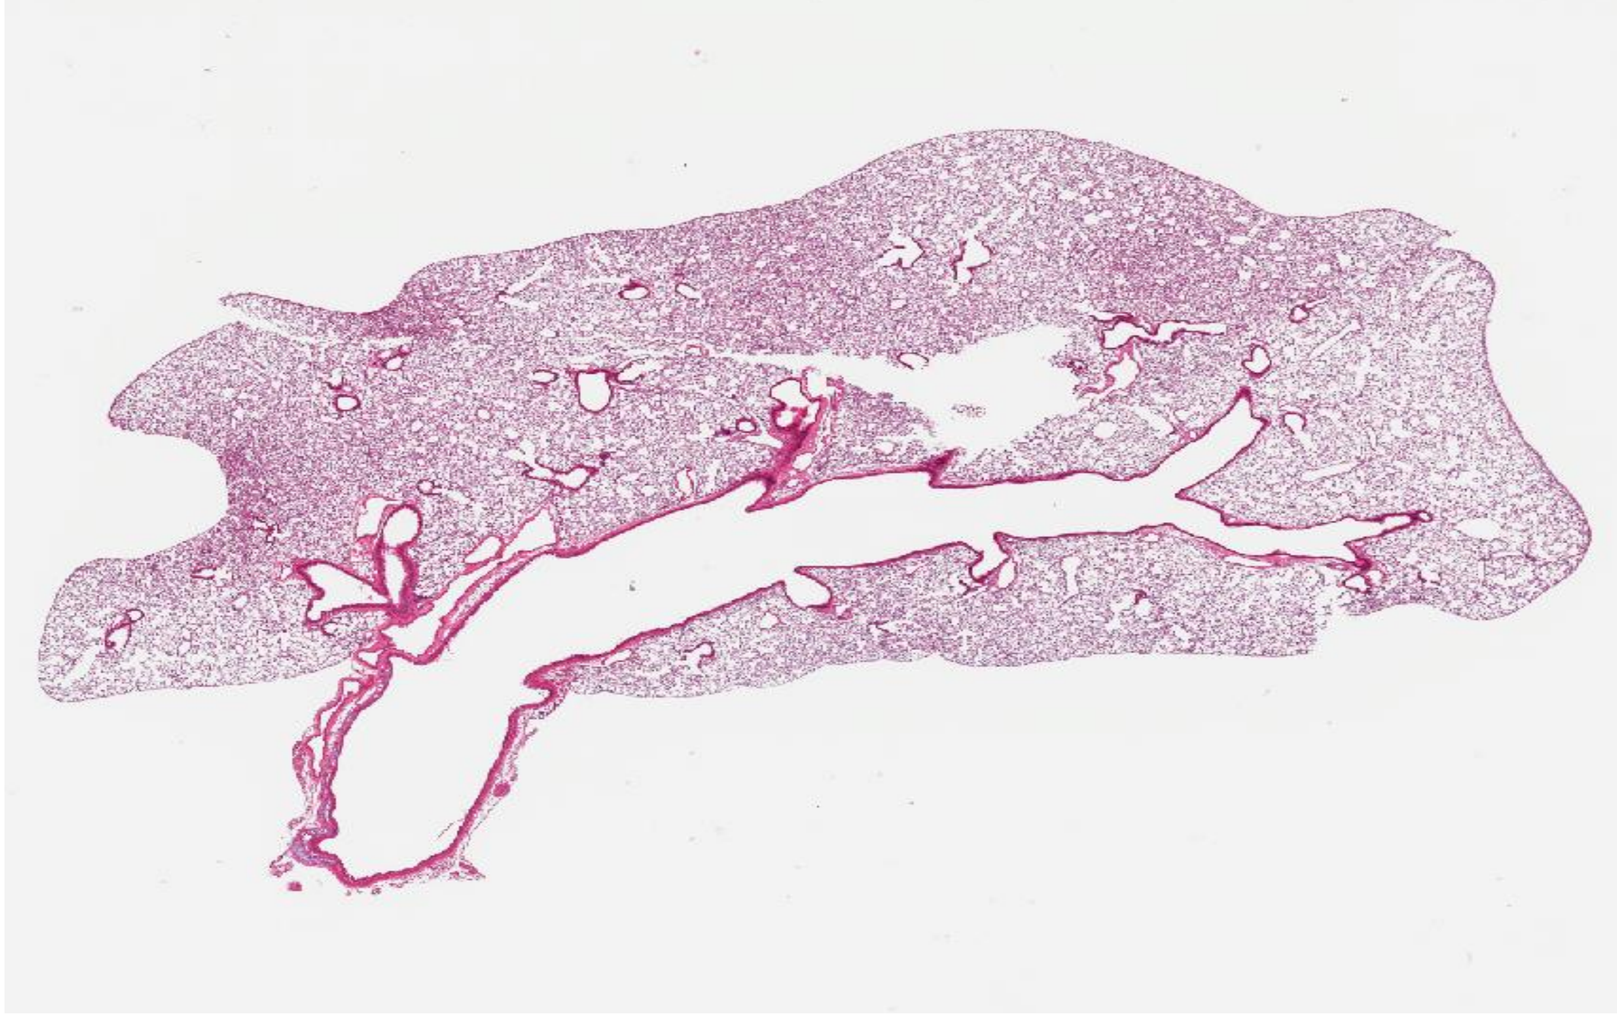
Fig. 1- 1 week 70% ethanol fixation

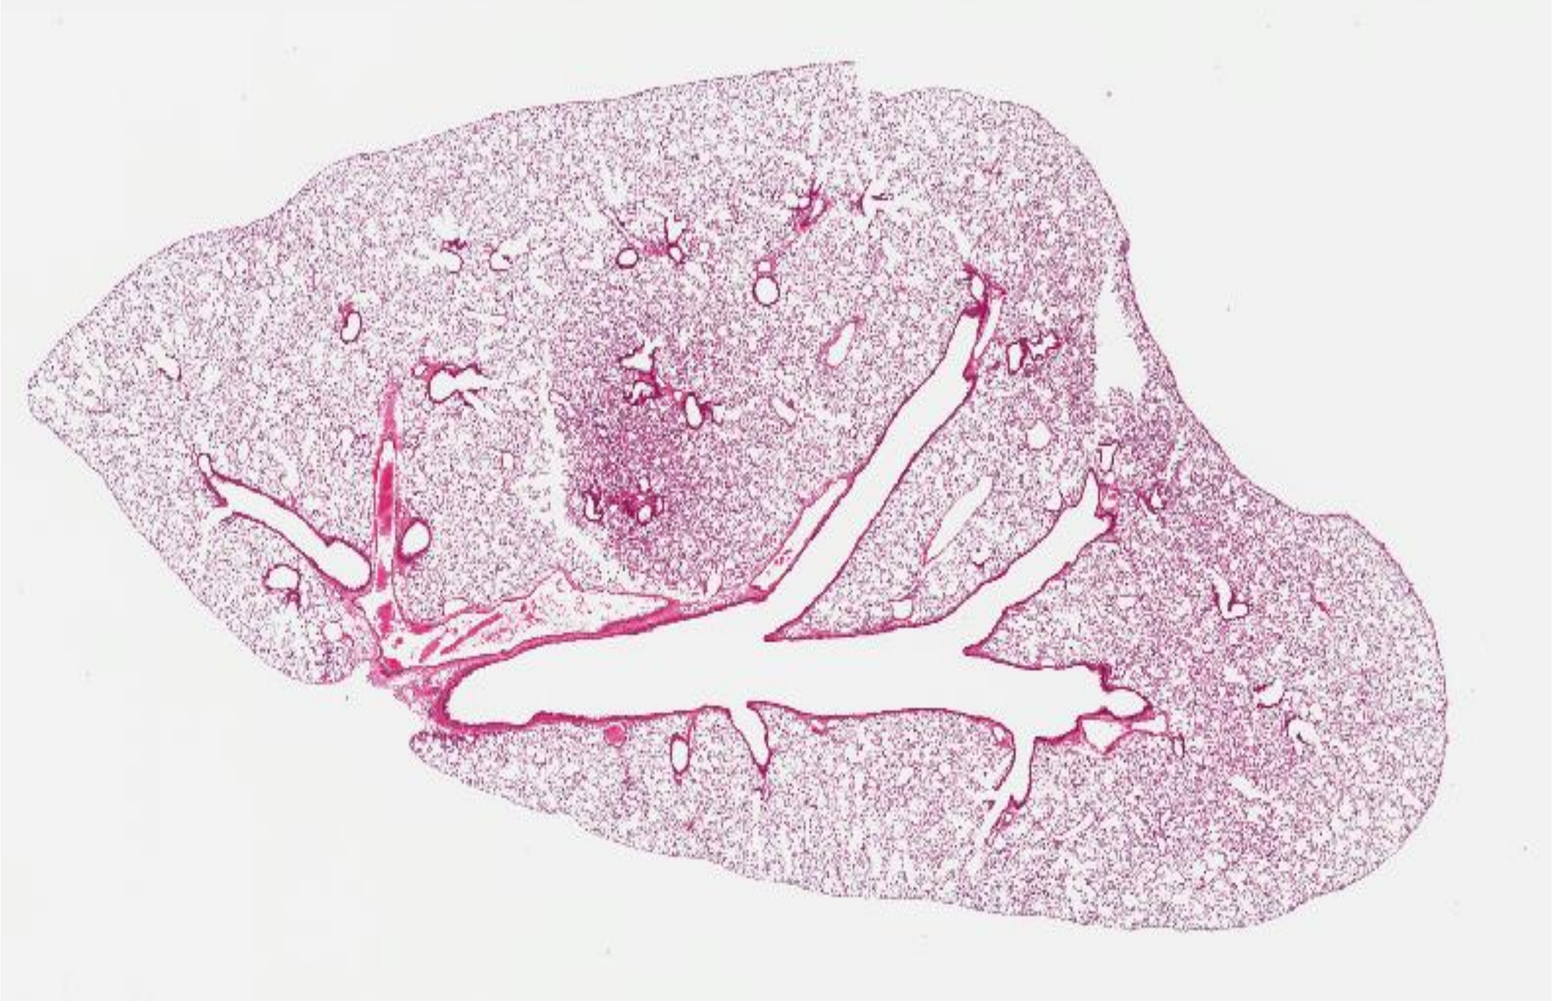
Fig. 2- 3 weeks 70% ethanol fixation

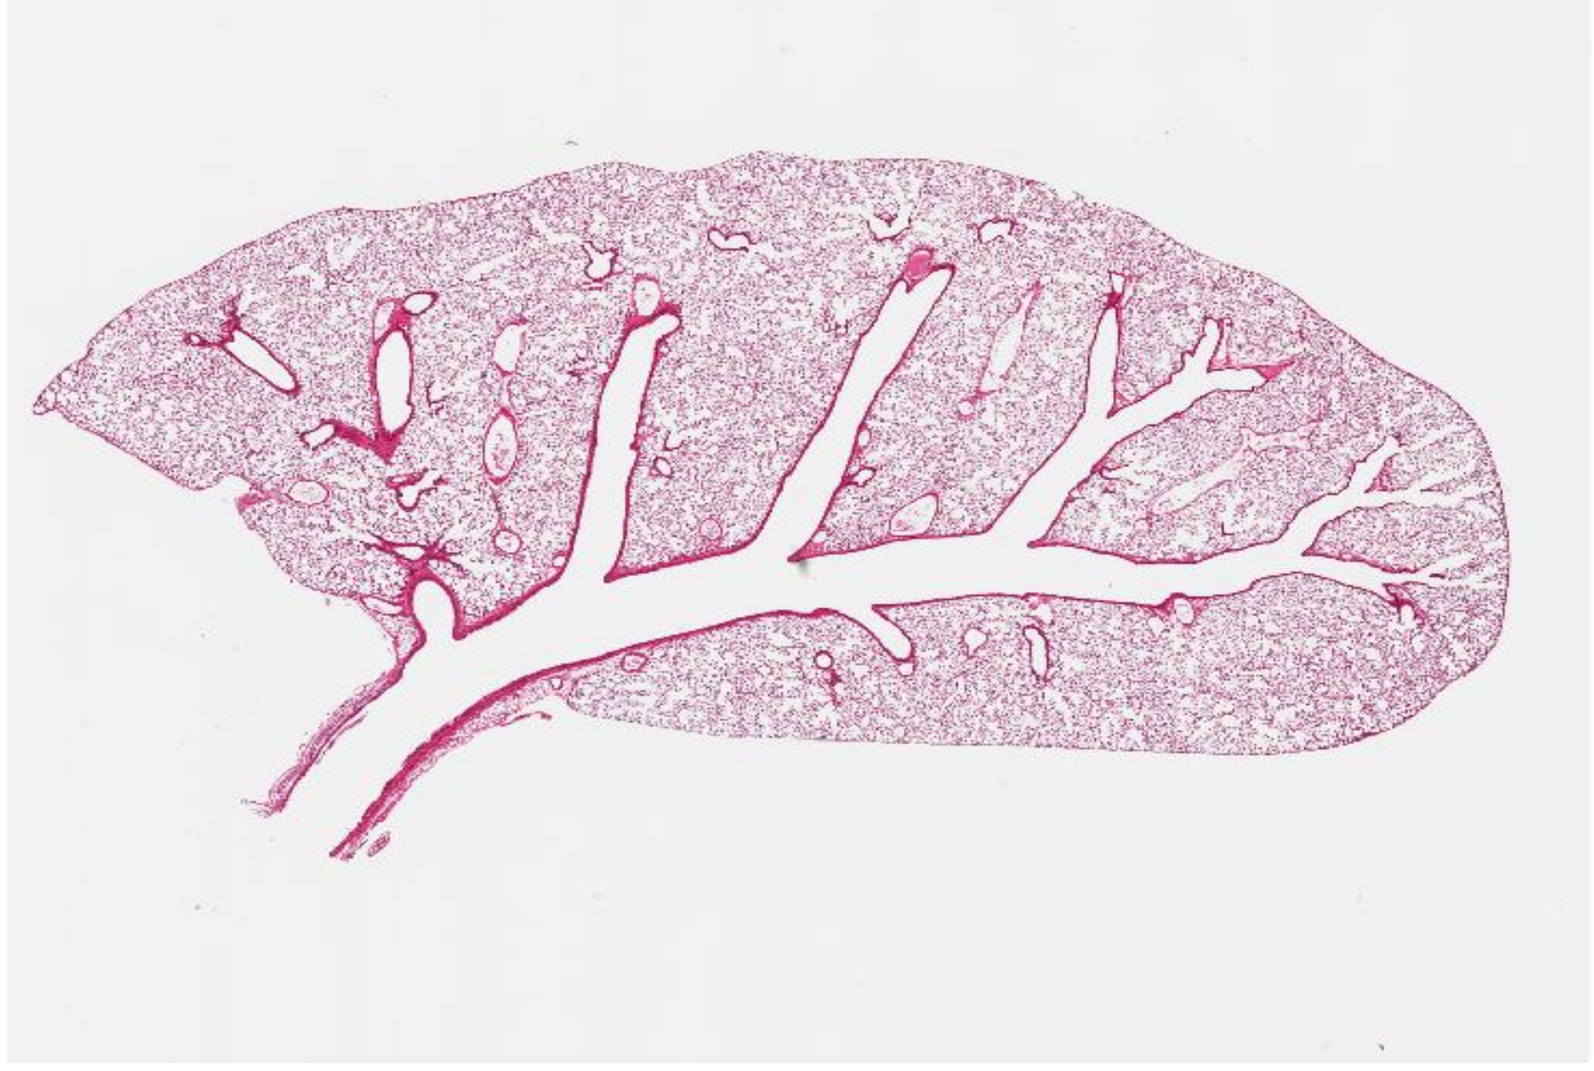
Fig. 3- 6 weeks 70% ethanol fixation

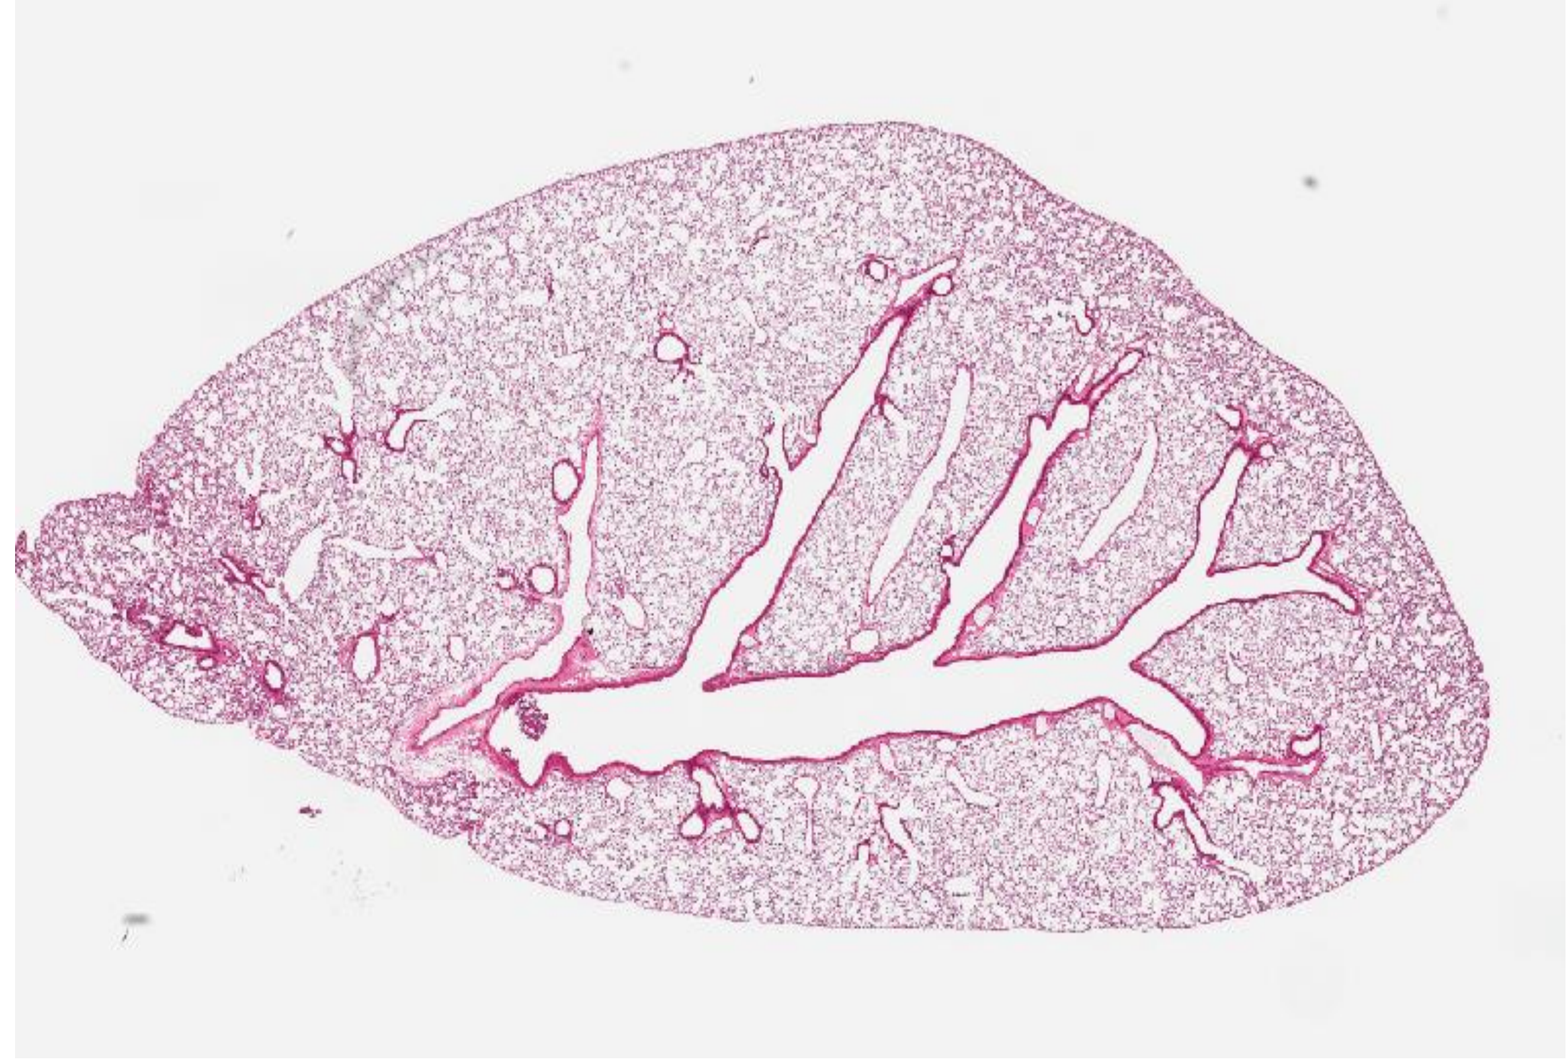
Fig. 4- 12 weeks 70% ethanol fixation

Results

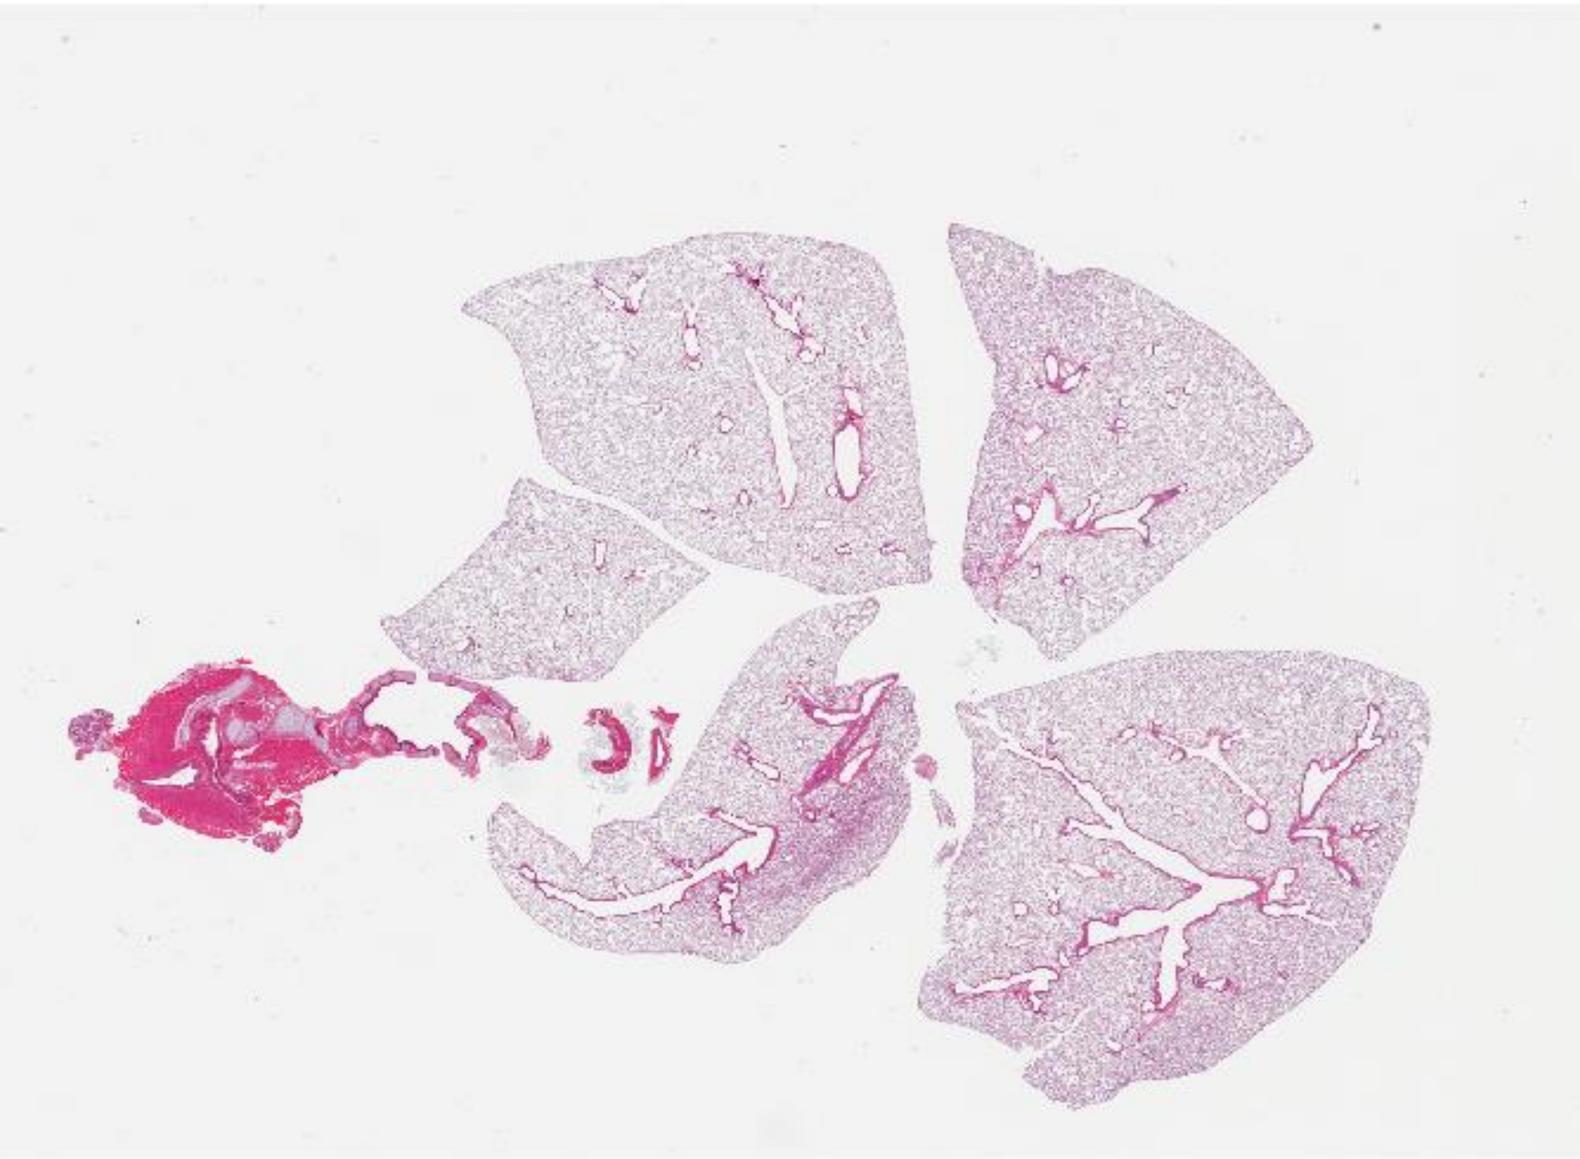
Fig. 5- 1 week 4 % formaldehyde fixation

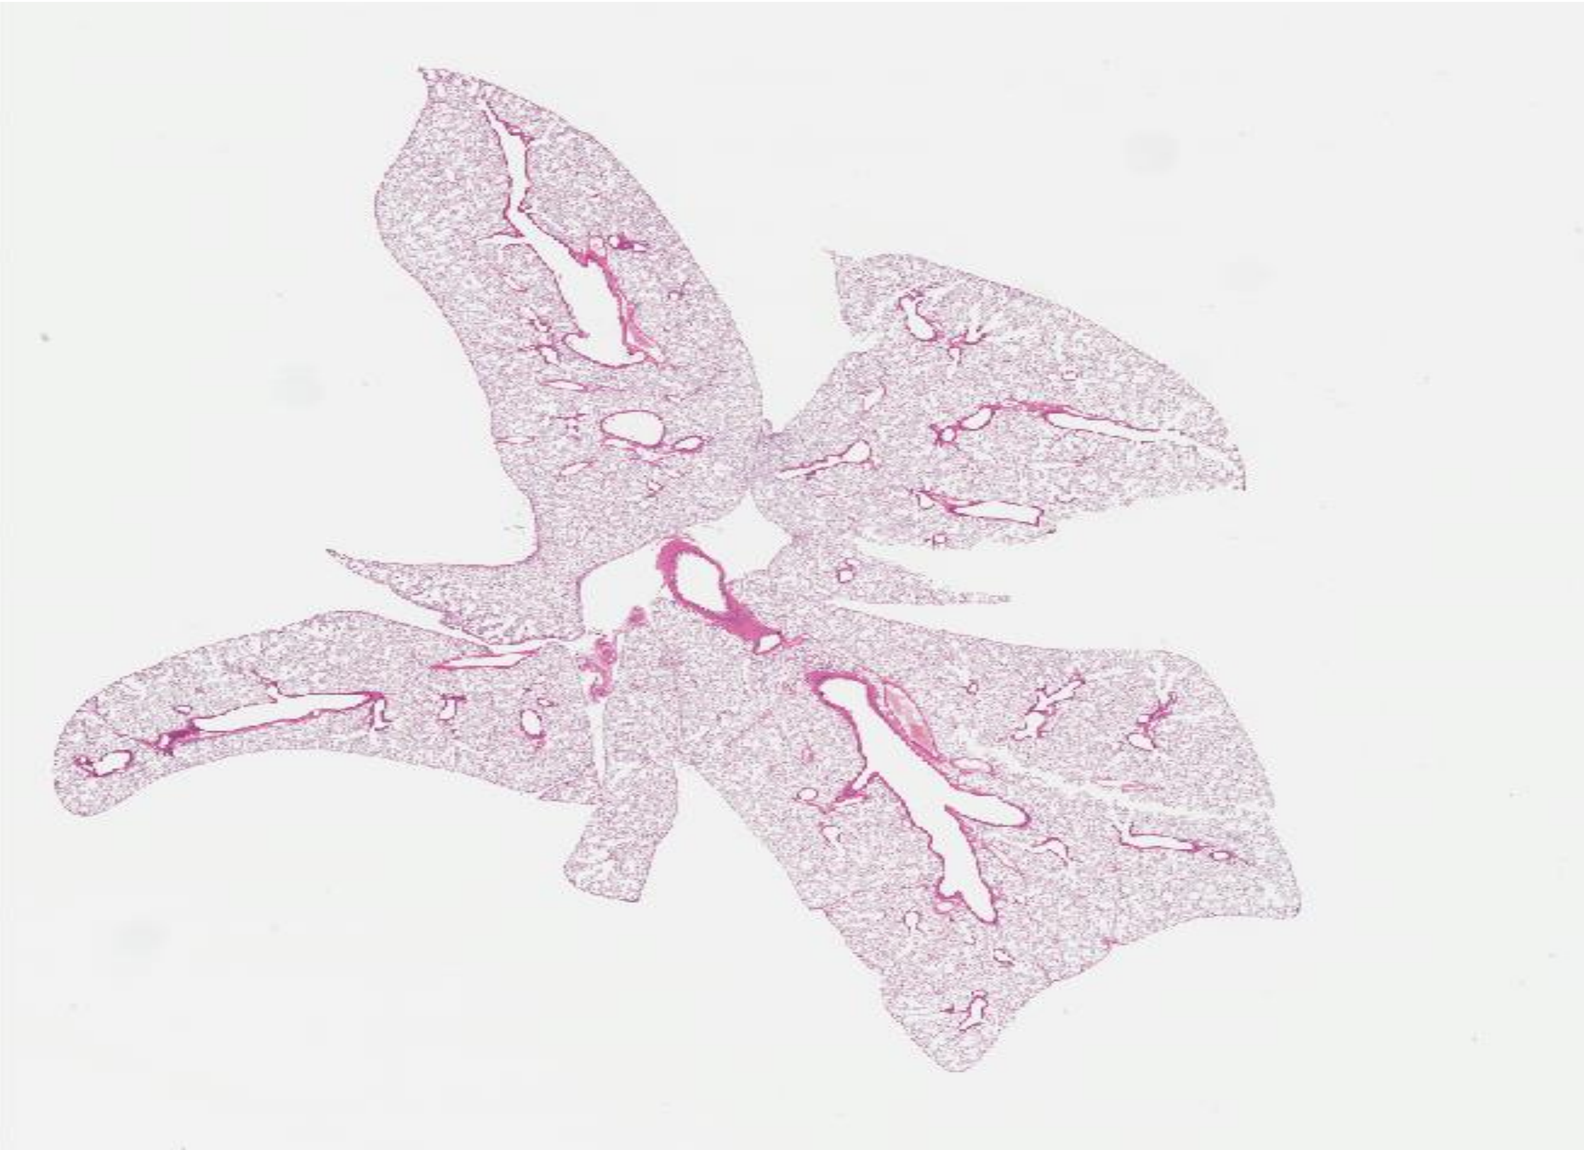
Fig. 6- 12 weeks 4% formaldehyde fixation

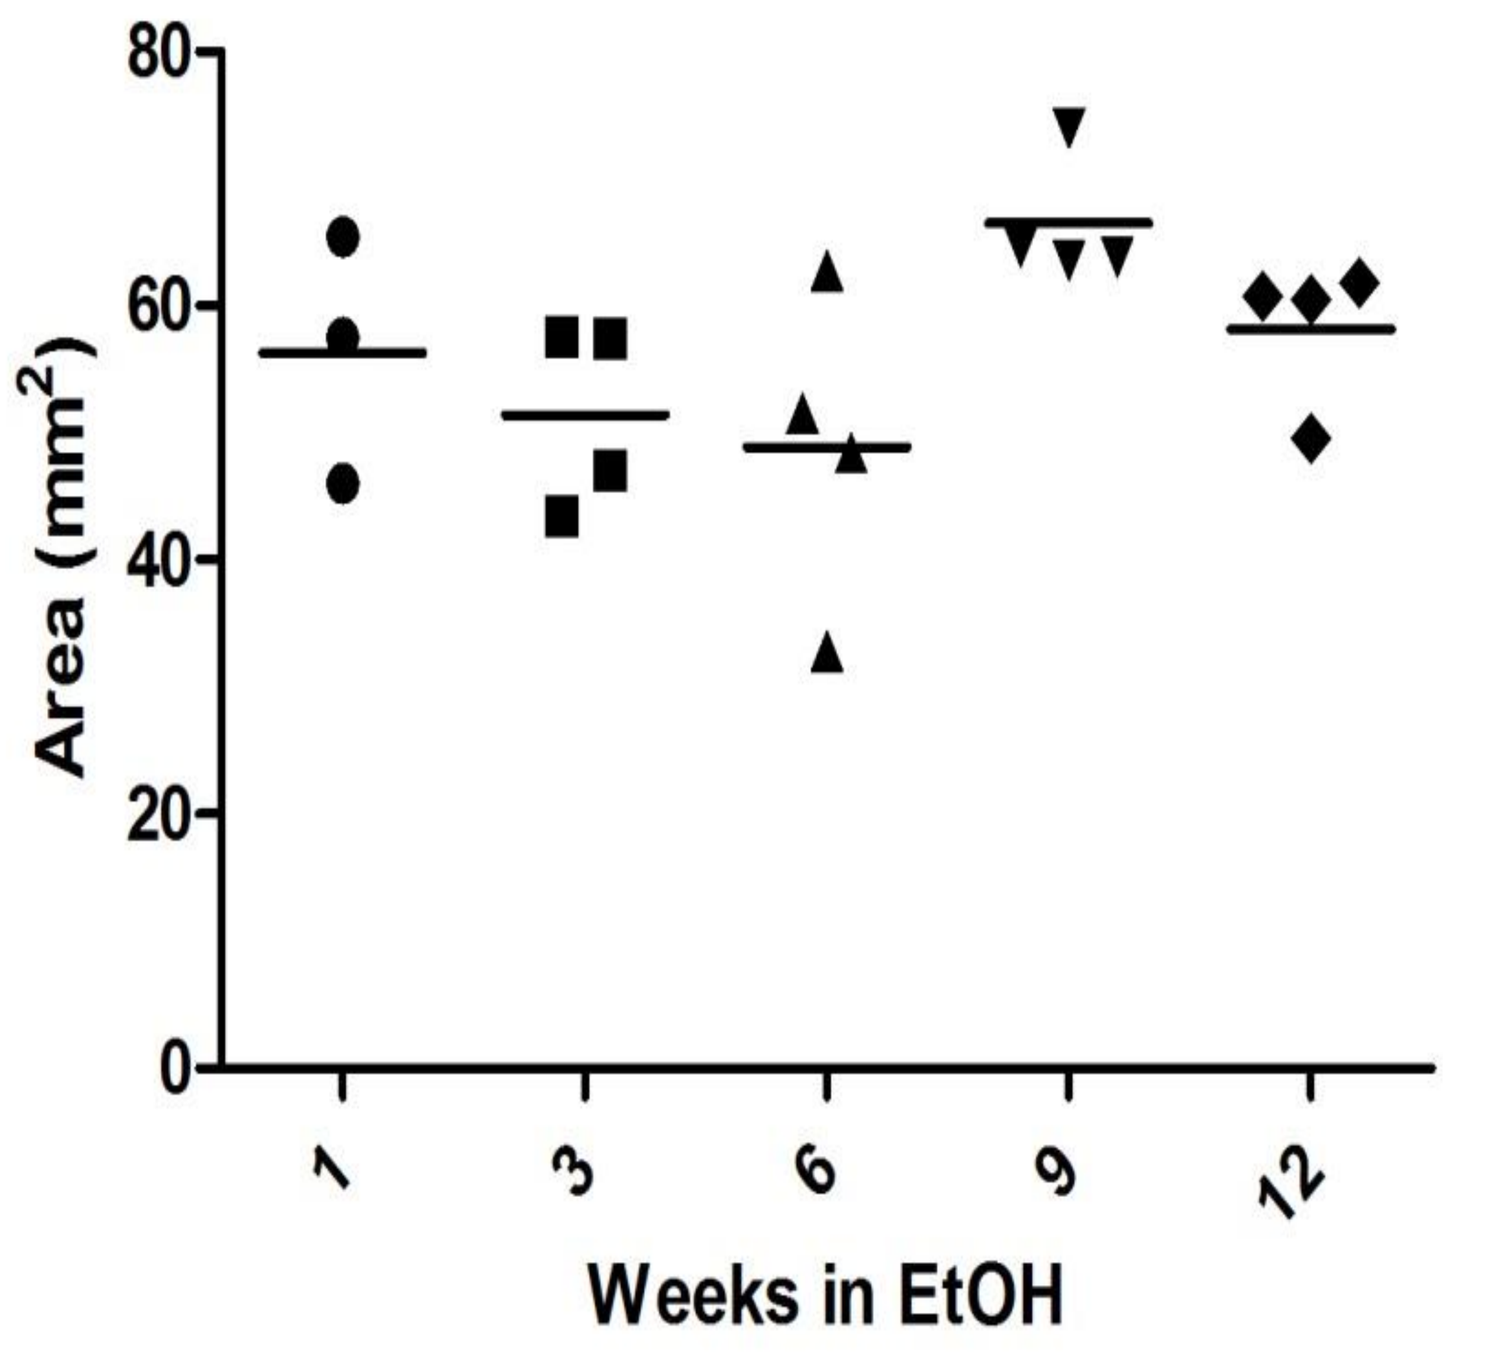
Area (mm2)
0
20
40
60
80
1
3
6
9
12
Weeks in EtOH
Fig. 7- Dimension of left lung with fixation in 70% ethanol

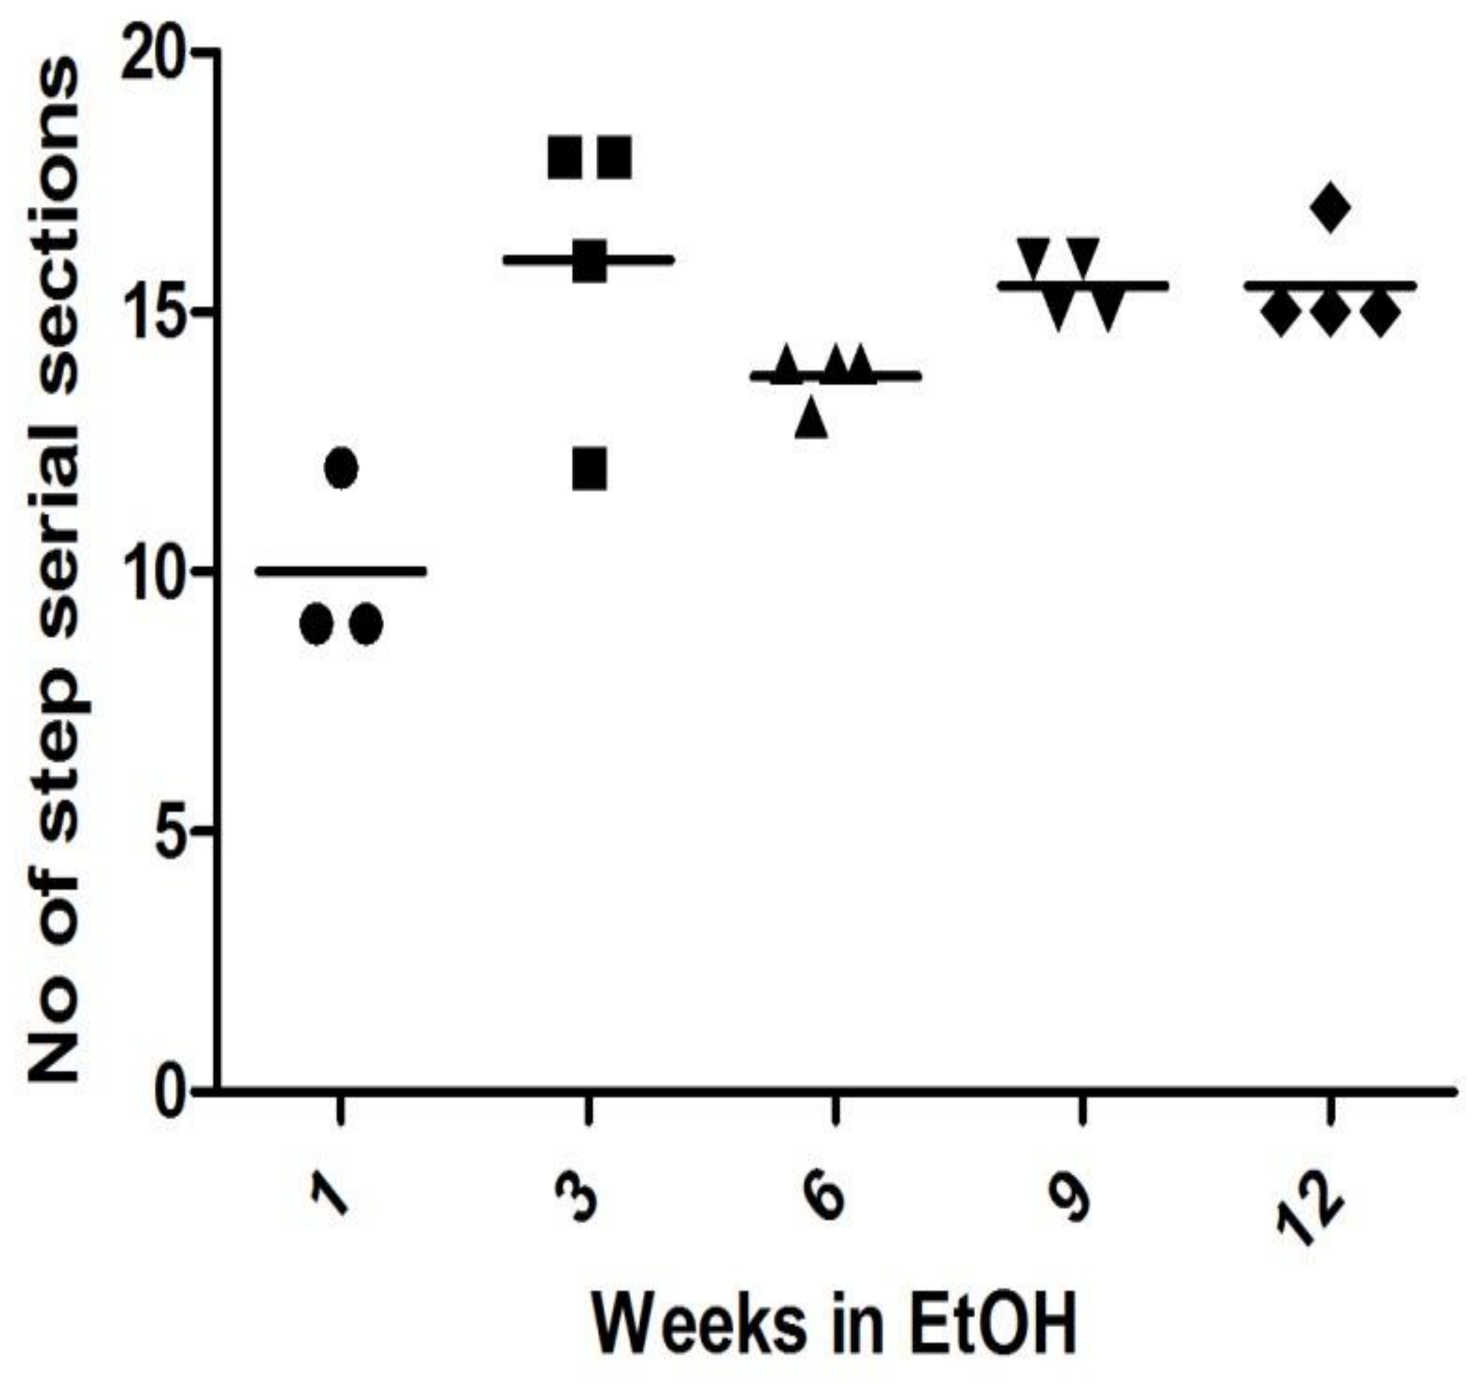
No of step serial sections
0
5
10
15
20
1
3
6
9
12
Weeks in EtOH
Fig. 8- No of step serial sections of left lung with fixation in 70% ethanol

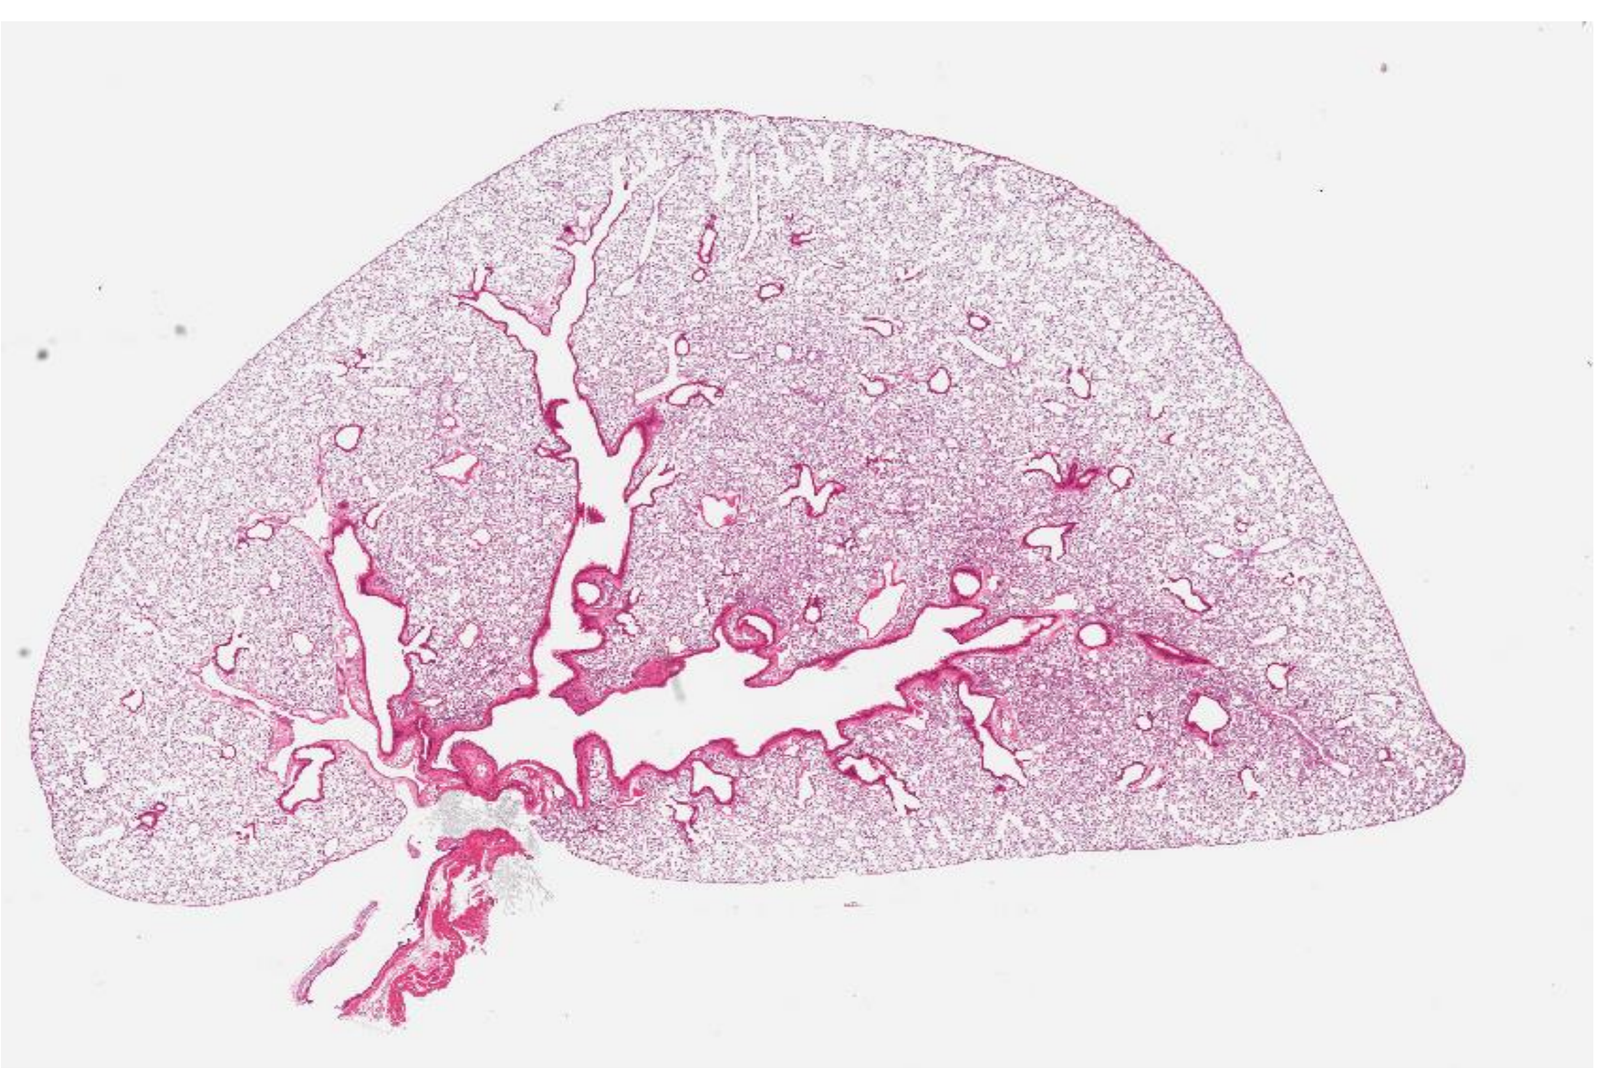
Fig. 9- Left lung embedded with compression.

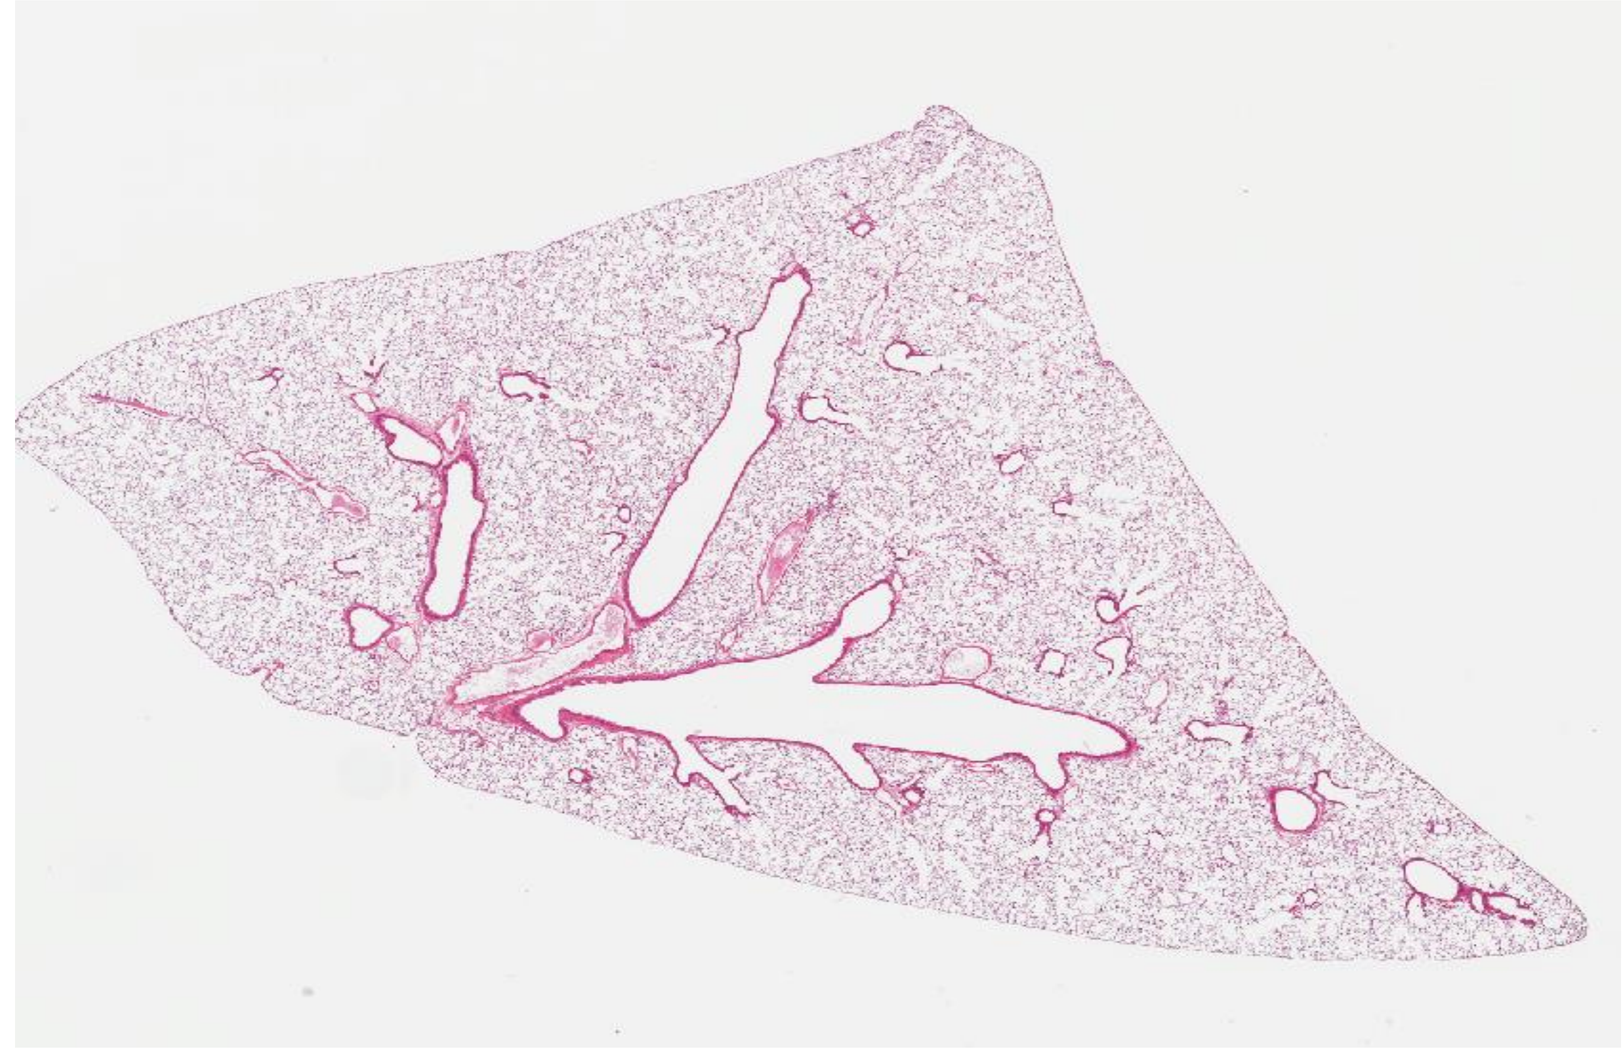
Fig. 10-Left lung embedded without deliberate compression.

Conclusions

- The tissue artifacts observed were related to neither the duration of storage of lungs prior to embedding, nor to the fixation solution used;
- Tissue artifacts seen in previous *in vivo* were reconstructed from lung embedded with deliberate compression;
- Physical compression of tissue including the use of tweezers during the embedding process must be avoided when handling murine lung tissues.

References

1. National Advisory Committee for Laboratory Animal Research (NACLAR), Guidelines on the care and use of animals for scientific purposes. Singapore, 2004
2. Braber S. et al., A comparison of fixation methods on lung morphology in a murine model of emphysema. Am J Physiol Lung Cell Mol Physiol 299, 2010
3. Theory and Pratice of Histological Techniques, Bancroft and Gamble, 6th edition 2008 Churchill Livingstone

Acknowledgement

The authors wish to thank all technical staff for their excellent work and Histovia Toxicologic Pathology for the histopathological evaluation.

PMI RESEARCH & DEVELOPMENT

CHART YOUR COURSE nsh 2013 NSH SYMPOSIUM CONVENTION Providence, RI September 20-25, 2013

Philip Morris International Research & Development, Quai Jeanrenaud 5, 2000 Neuchâtel, Switzerland
T: +41 58 242 21 11, **F**: +41 58 242 28 11, **W**: www.philipmorrisinternational.com