A Mechanistic Study of Cigarette Smoke-Induced COPD in C57BL/6 Mice: The changes in lung epigenome following smoking cessation or switching to aerosol from a prototypic modified risk tobacco product

Sierro N.[1], Talikka M.[1], Hoeng J.[1], Peitsch MC.[1], Hayes, AW.[2], Ivanov, NV.[1]
[1]Philip Morris International R&D, Philip Morris Products S.A., 2000 Neuchatel, Switzerland.
[2]Spherix Consulting, Rockville, MD, USA and Scientific Advisory Board PMI R&D.

2260

Introduction

Chronic Obstructive Pulmonary Disease (COPD) is the major cause of chronic morbidity and mortality in the world[1]. It is defined as progressive, irreversible airflow limitation caused by the combined effects of emphysema, chronic bronchitis, and narrowing of the small airways[2]. In the western world cigarette smoke is the main etiological factor in the pathogenesis of COPD[3]. Even though aberrant histone modifications have been the focus of epigenetic studies around COPD[4], some studies have indicated that DNA methylation might also play a role in the COPD pathology. p16 and GATA4 methylation has been associated with lower percent predicted FEV1 in sputum[5]. A larger scale analysis from white blood cells proposed DNA methylation as a possible biomarker of COPD. 70% of the identified 349 methylated CpG sites were outside of CpG islands and significantly associated with COPD severity[6]. In addition to the predictive value, such methylation patterns may identify new molecular pathways involved in COPD pathogenesis. Known mechanisms by which cigarette smoke modulates DNA methylation have been reviewed by Lee and Pausova[7,8], and are shown in figure 1.

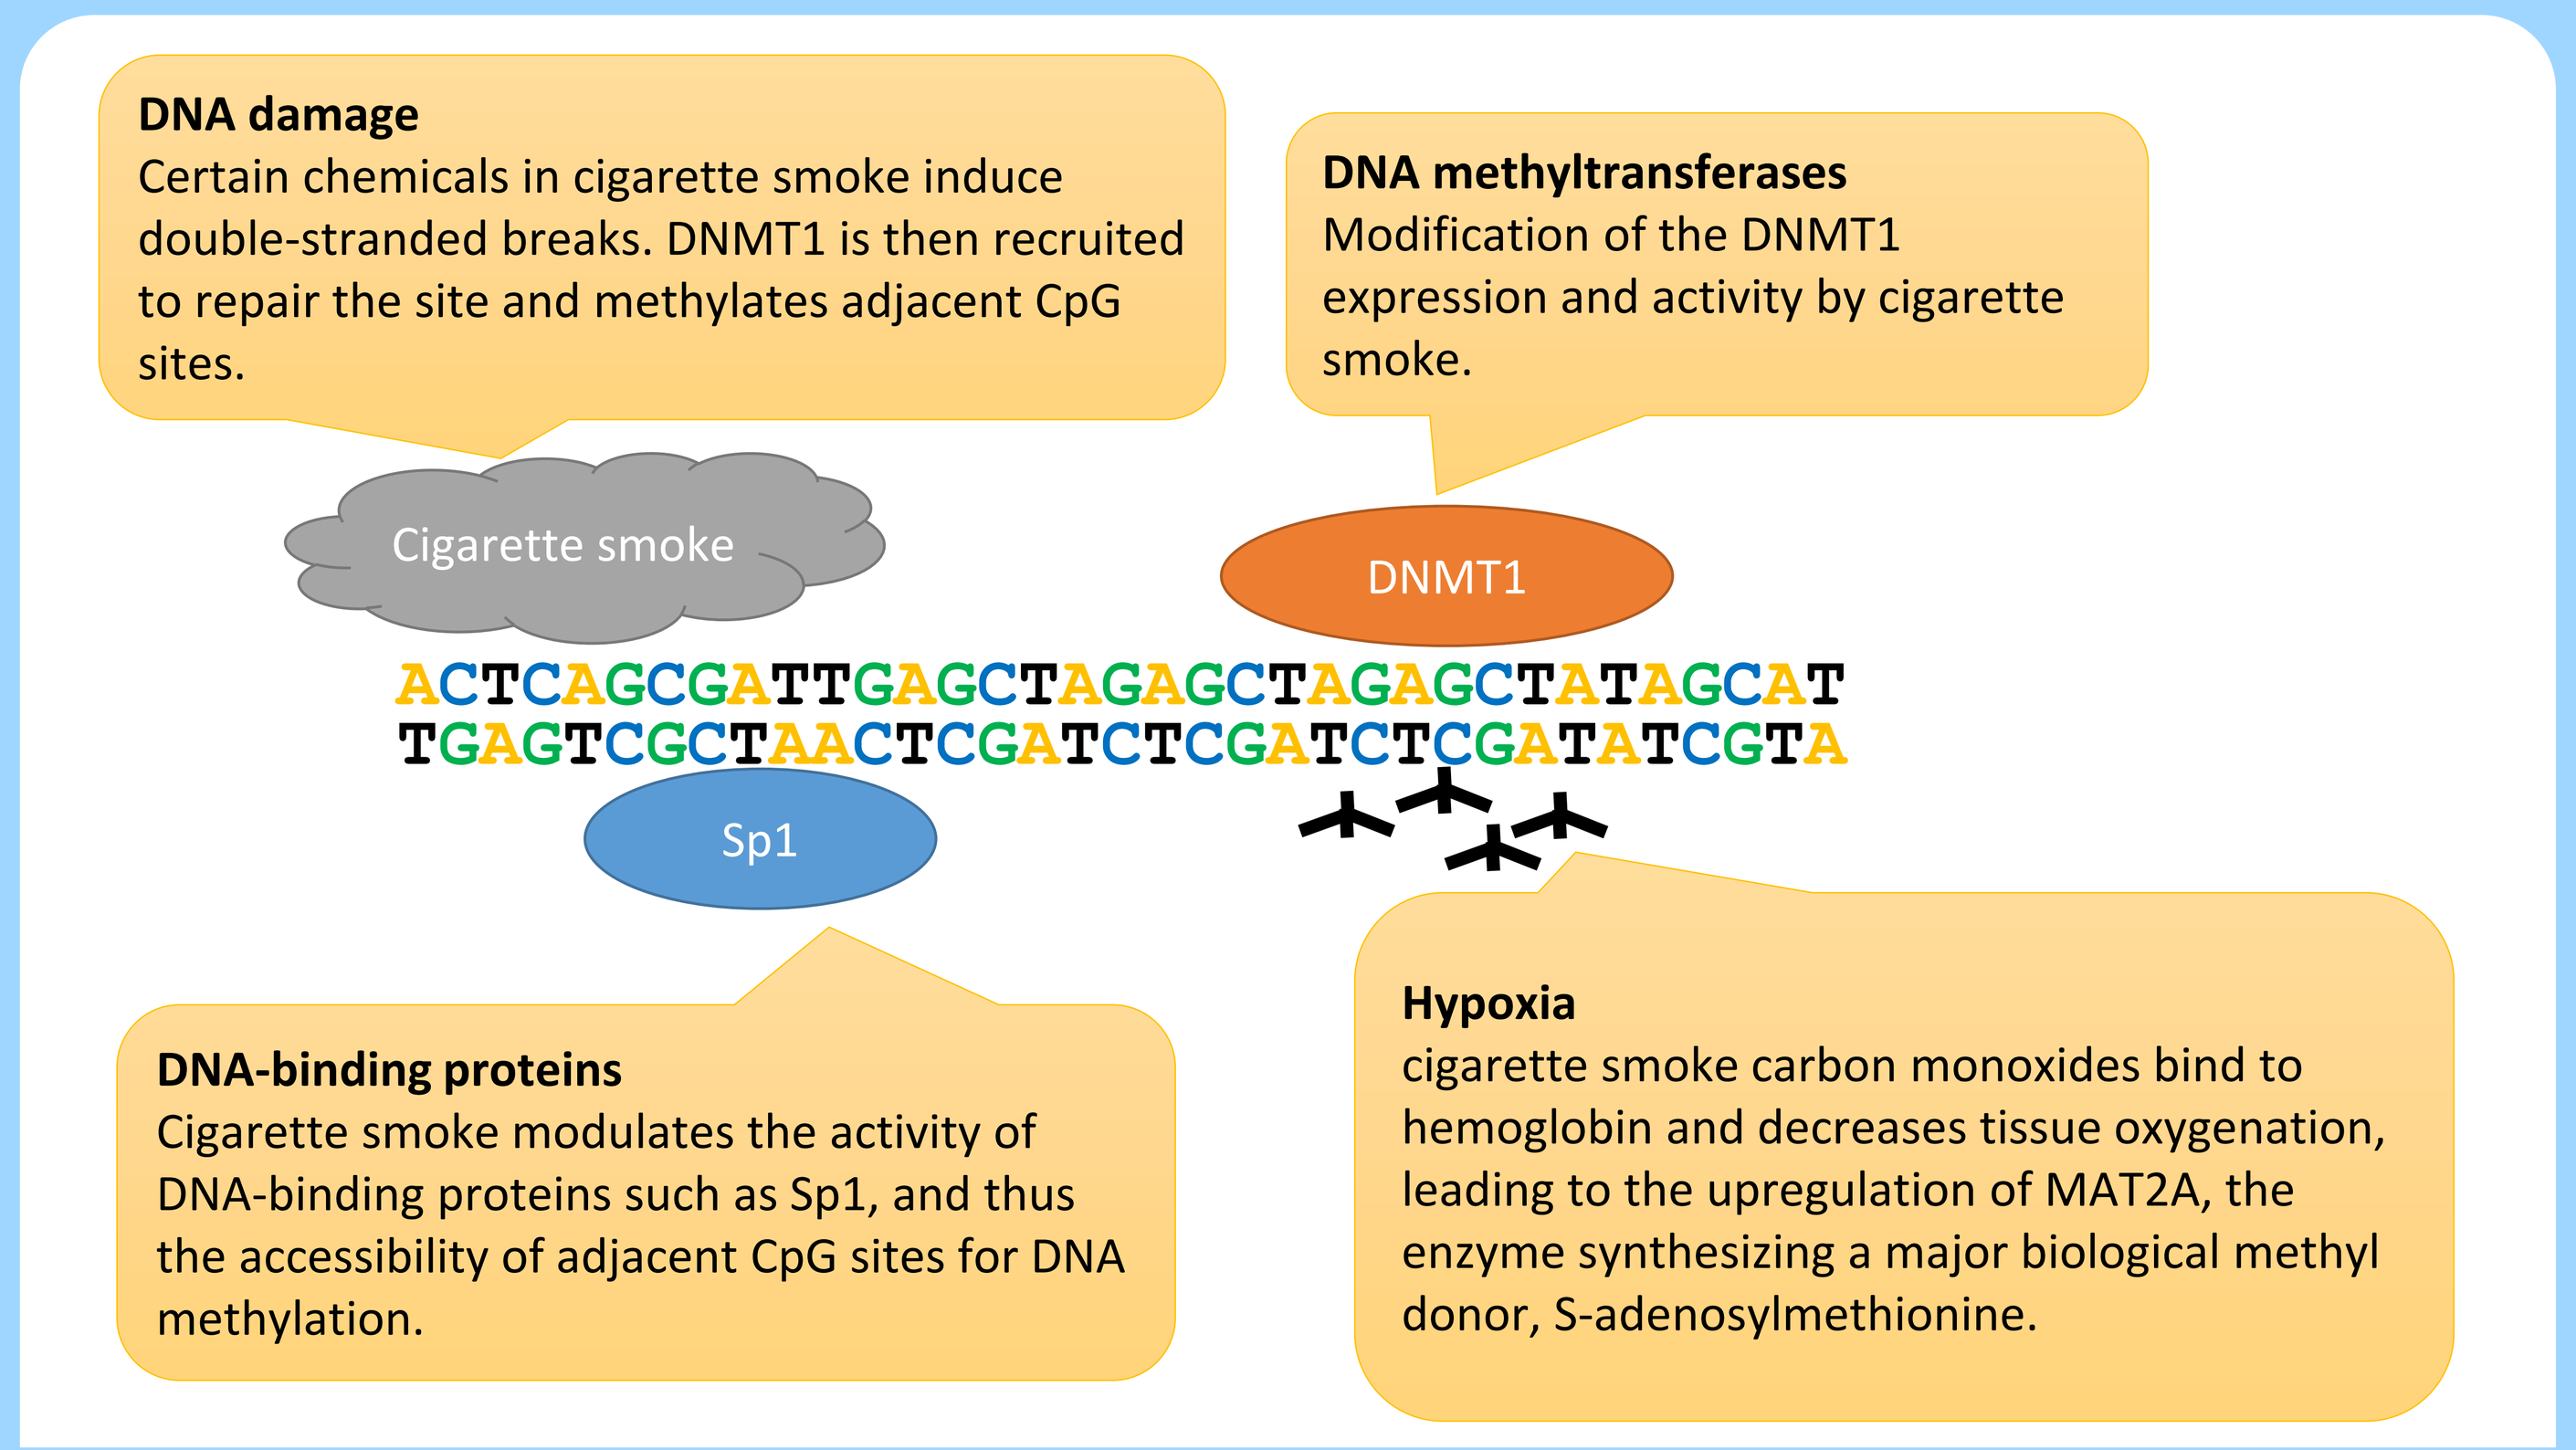
DNA damage
Certain chemicals in cigarette smoke induce double-stranded breaks. DNMT1 is then recruited to repair the site and methylates adjacent CpG sites.
DNA methyltransferases
Modification of the DNMT1 expression and activity by cigarette smoke.
Cigarette smoke
DNMT1
ACTCAGCGATTGAGCTAGAGCTAGAGCTATAGCAT
TGAGTCGCTAACTCGATCTCGATCTCGATATCGTA
Sp1
DNA-binding proteins
Cigarette smoke modulates the activity of DNA-binding proteins such as Sp1, and thus the accessibility of adjacent CpG sites for DNA methylation.
Hypoxia
cigarette smoke carbon monoxides bind to hemoglobin and decreases tissue oxygenation, leading to the upregulation of MAT2A, the enzyme synthesizing a major biological methyl donor, S-adenosylmethionine.

Figure 1: Cigarette smoke and DNA methylation.
Cigarette smoke can modulate DNA methylation by inducing DNA damage, modifying the expression of genes encoding DNA methyltransferases and DNA-binding proteins and/or the activity of the proteins, and by reducing tissue oxygenation[7].

Among various mouse models for the study of experimental emphysema/COPD, it has been shown that C57BL/6 mouse is a useful tool for cigarette smoking-induced COPD studies[9]. Even though the model mimics only some aspects of early human COPD, characterized by reduced lung function, abnormal inflammatory response in the airways, small airway remodeling, and the destruction of lung alveolar tissue[10,11], it provides valuable insights into emphysema initiation and progression[9].

In this study, the impact of inhalation of aerosol from a reference cigarette (3R4F) or a prototypic modified risk tobacco product (pMRTP), was evaluated in the lung methylome of C57BL/6 mice. Mice were exposed to aerosol from 3R4F, pMRTP or filtered air for up to 7 months. After 2 months of exposure to 3R4F, switching and cessation groups were exposed to pMRTP or filtered air, respectively.

References

1. Mathers, C.D. and Loncar, D. (2006), *PLoS Med*, **3**, e442.
2. MacNee, W. (2006), *BMJ: British Medical Journal*, **332**, 1202.
3. Pauwels, R.A., *et al.* (2001), Am J Respir Crit Care Med, **163**, 1256-1276.
4. Yang, I.A., *et al.* (2011), *Expert Opin Ther Targets*, **15**, 439-456.
5. Sood, A., *et al.* (2010), *Am J Respir Crit Care Med*, **182**, 1098-1104.
6. Qiu, W., *et al.* (2012), *Am J Respir Crit Care Med*, **185**, 373-381.
7. Lee, K.W. and Pausova, Z. (2013), *Front Genet,* **4**, 132.
8. Talikka, M. *et al.* (2012), *Crit Rev Toxicol,* **10**, 877-889.
9. Churg, A., Cosio, M. and Wright, J.L. (2008), *Am J Physiol Lung Cell Mol Physiol*, **294**, L612-L631.
10. Pauwels, R.A. and Rabe, K.F. (2004), *Lancet*, **364**, 613-620.
11. Rabe, K.F., *et al.* (2007), *Am J Respir Crit Care Med*, **176**, 532-555.
12. Dolzhenko, E. and Smith, A.D. (2014), *BMC Bioinformatics*, **15**, 215.
13. Love, M.I., Huber, W. and Anders, S. (2014), *Genome Biology*, **15**, 550.

Results and Conclusion

Whole genome DNA methylation

The DNA isolated from mouse lungs was treated with bisulfite to convert unmethylated cytosines to uracil. After paired-end sequencing on an Illumina HiSeq2500 and mapping to the reference mouse genome, unmethylated cytosines from the first read were identified as C/T mismatches, and those from the second read as G/A mismatches (figure 2).

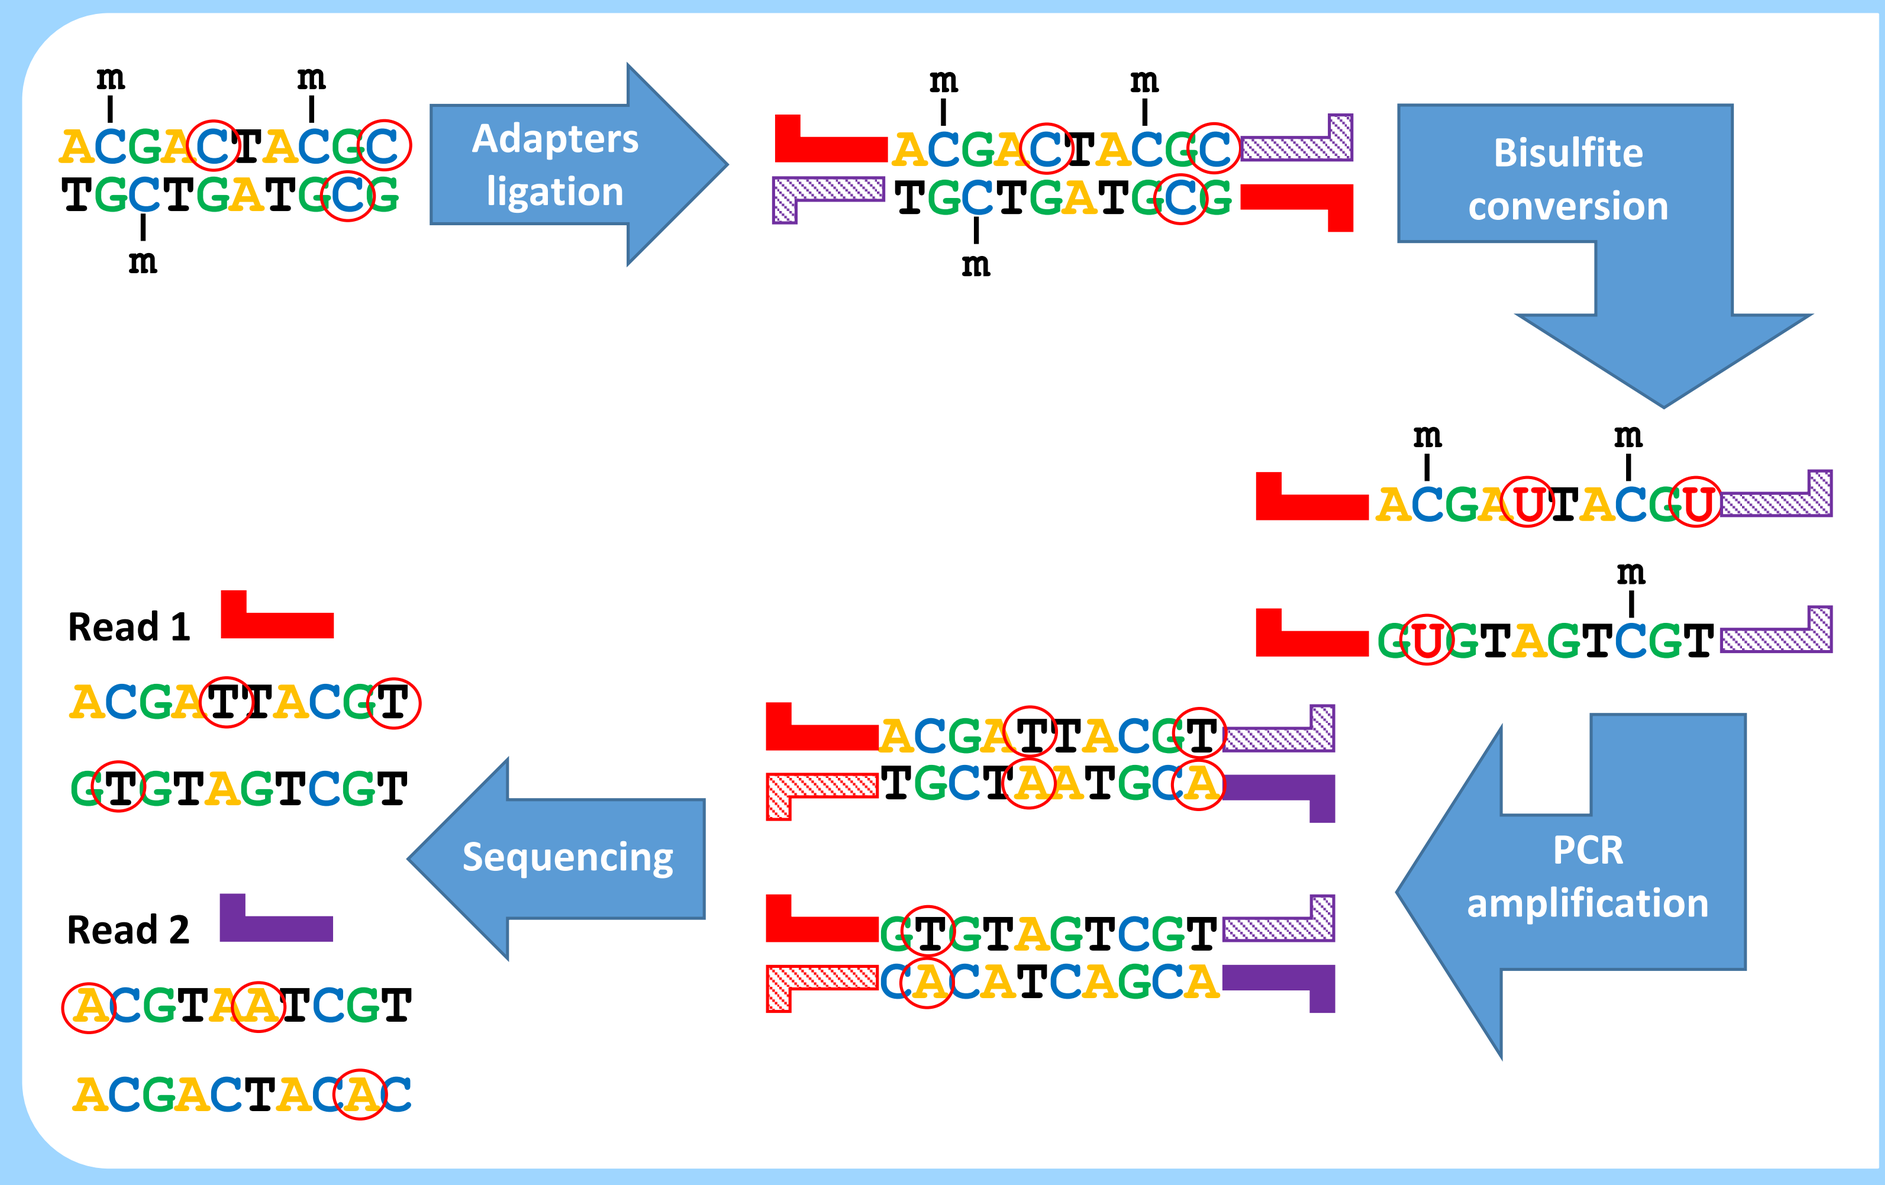
Adapters ligation
Bisulfite conversion
PCR amplification
Sequencing
Read 1
Read 2

Figure 2: DNA methylation sequencing.
Methylated sequencing adapters are ligated to DNA fragments before bisulfite conversion and PCR amplification.
Upon sequencing, the first read is therefore T-rich, and the second read A-rich. Red circles indicate originally unmethylated cytosines, and the corresponding sequenced nucleotide.

Methylation of CpG sites

Samples from the 2nd and 7th month of the study were sequenced at depth varying from 4X to 20X, resulting in methylation call for at least 70% of all the 21 million CpG loci in the mouse reference genome (figure 3). The average percentage of methylation for CpG sites outside for CpG islands was found to be around 75%, while that for CpG sites inside CpG islands was around 10% (figure 3). This is expected since promoters are enriched in CpG islands, whose methylation would result in a decrease of gene expression.

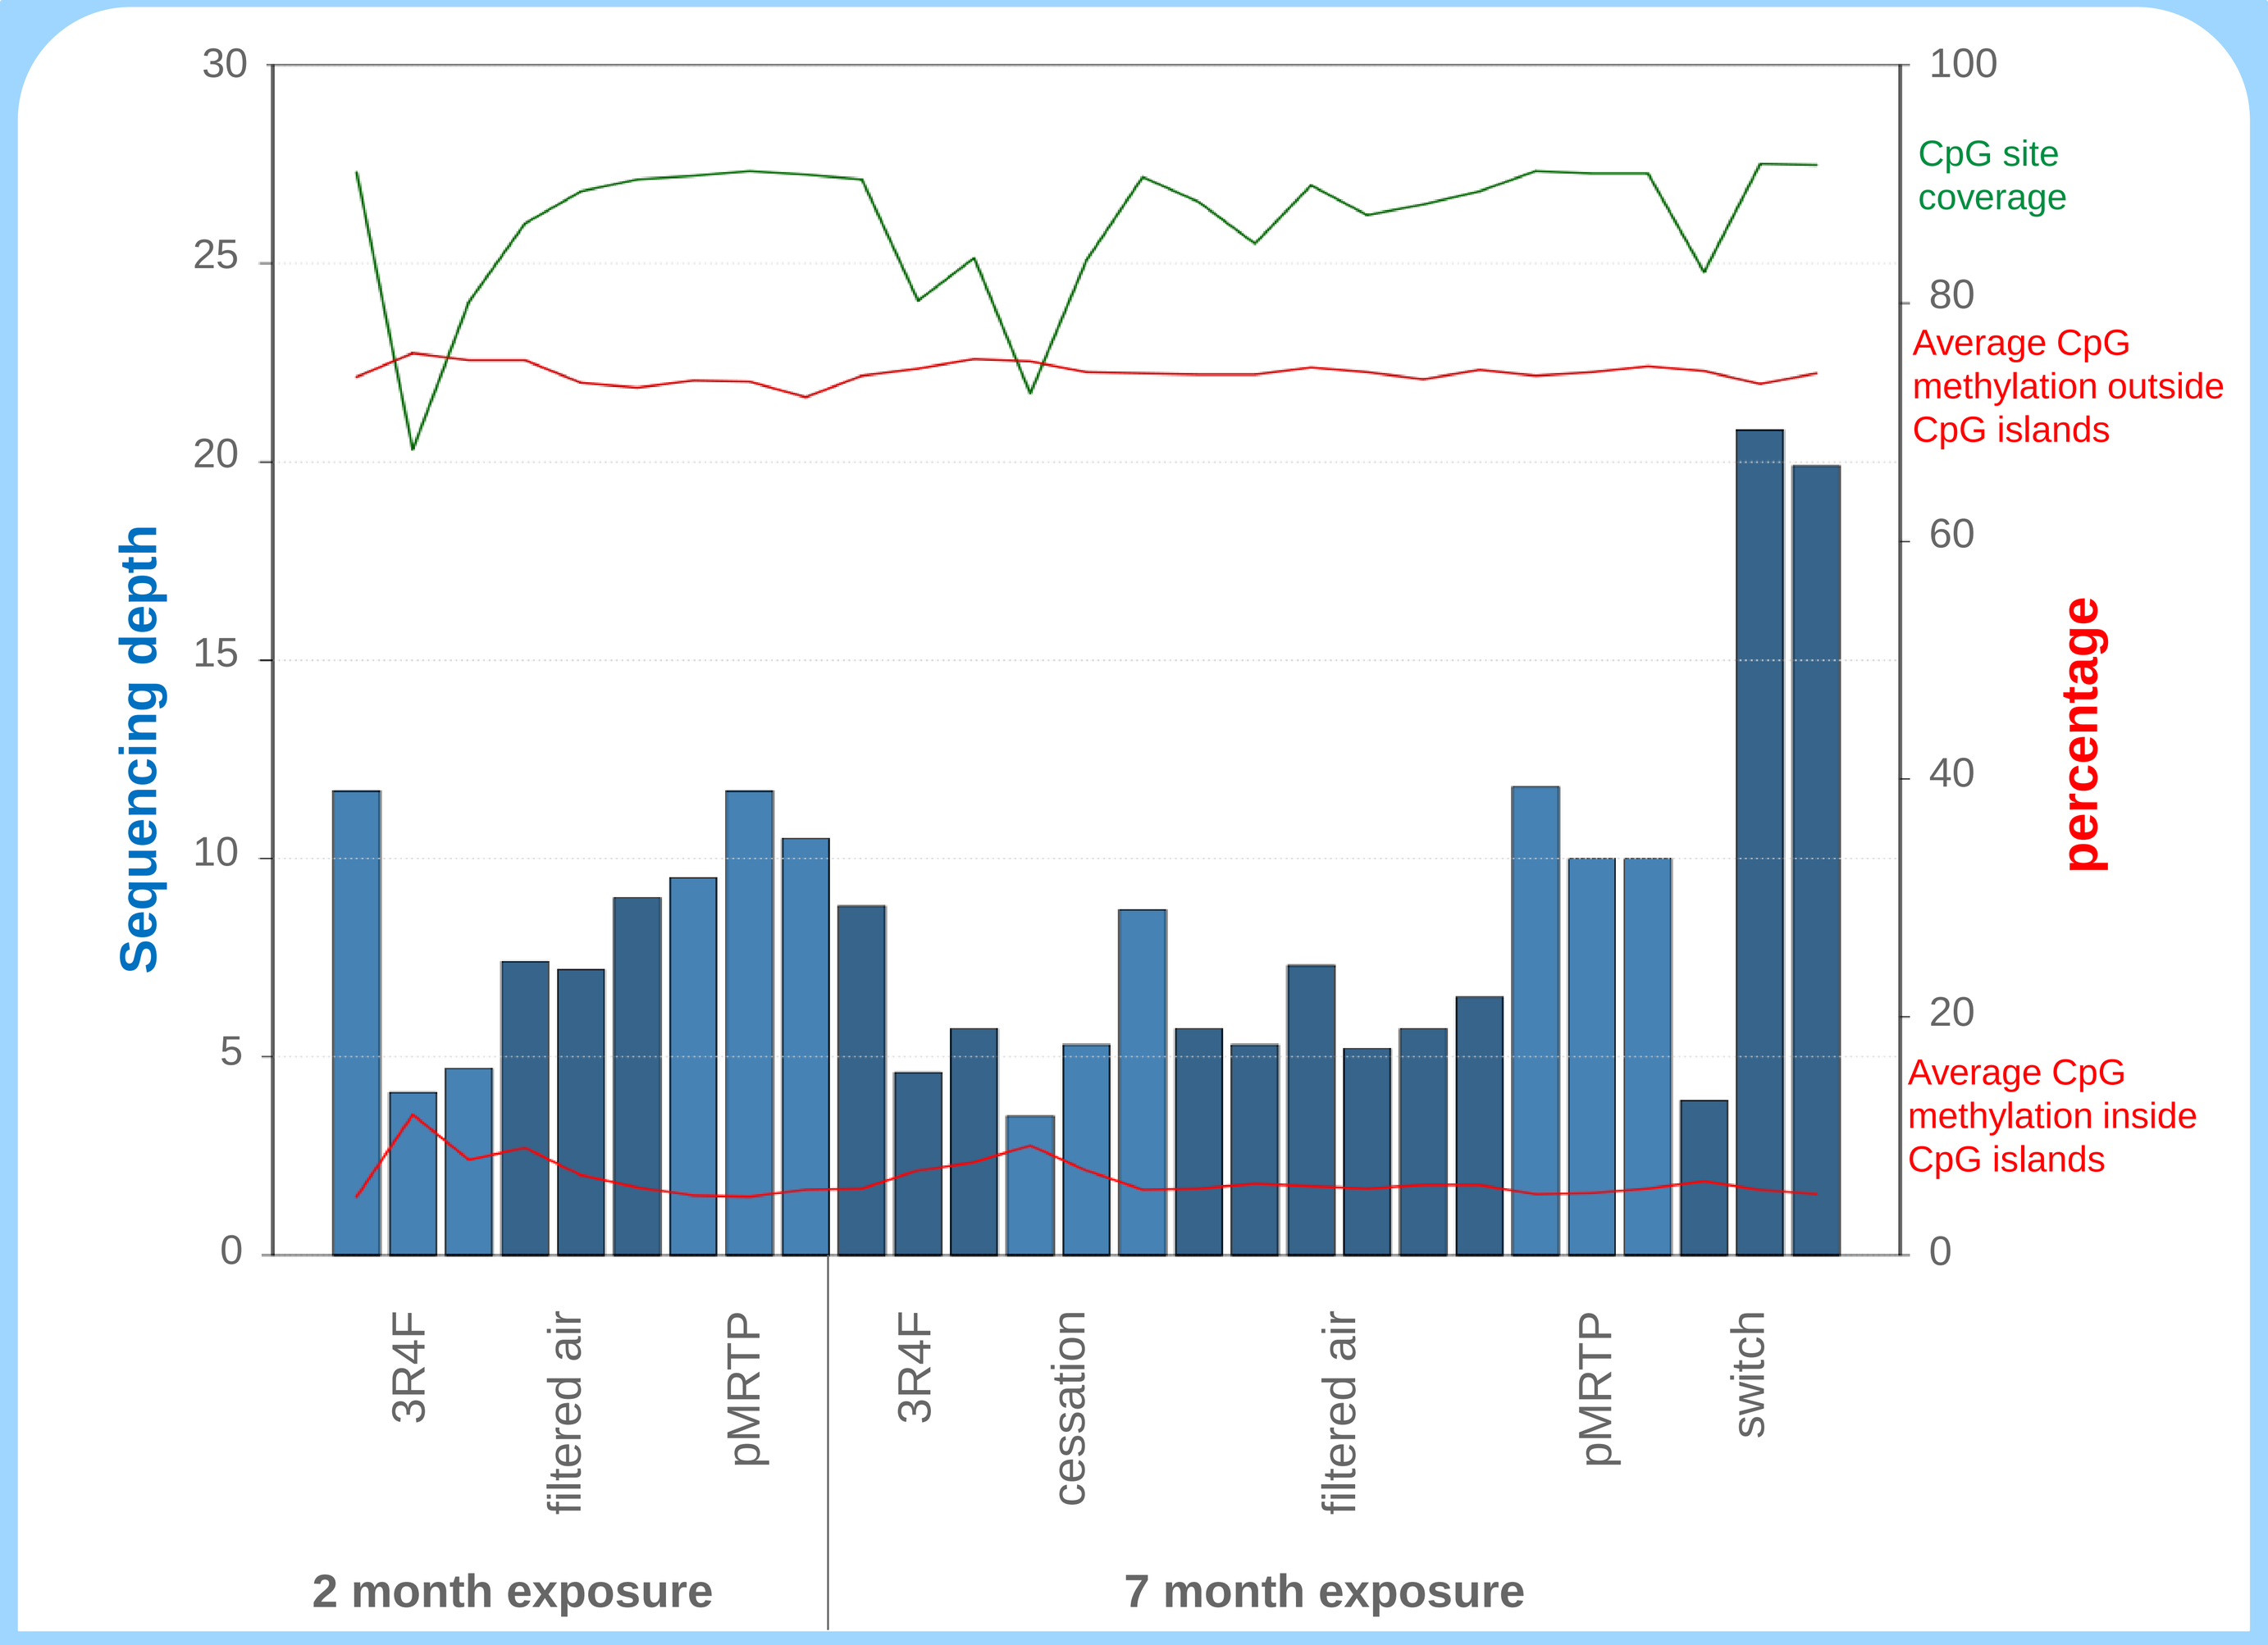
Sequencing depth
percentage
CpG site coverage
Average CpG methylation outside CpG islands
Average CpG methylation inside CpG islands
3R4F
filtered air
pMRTP
cessation
switch
2 month exposure
7 month exposure

Figure 3: Methylation of CpG sites.
Sequencing depth per samples is given by the blue bars. Light and dark blue are used to separate the different sample groups. The average percentages of DNA methylation of CpG sites inside and outside CpG islands are indicated by the red lines. The green line shows the percentage of CpG sites covered by methylation information.

Differentially methylated CpG sites

An increase in differentially methylated CpG sites upon continuous exposure to 3R4F aerosol was observed. Cessation resulted in a moderate increase of hypermethylated CpG sites and switching to a pMRTP to a decrease in the number of differentially methylated CpG sites. Exposure to a pMRTP aerosol showed after 2 months a large number of hypomethylated CpG sites, but very little differential methylation after 7 months (figure 4).

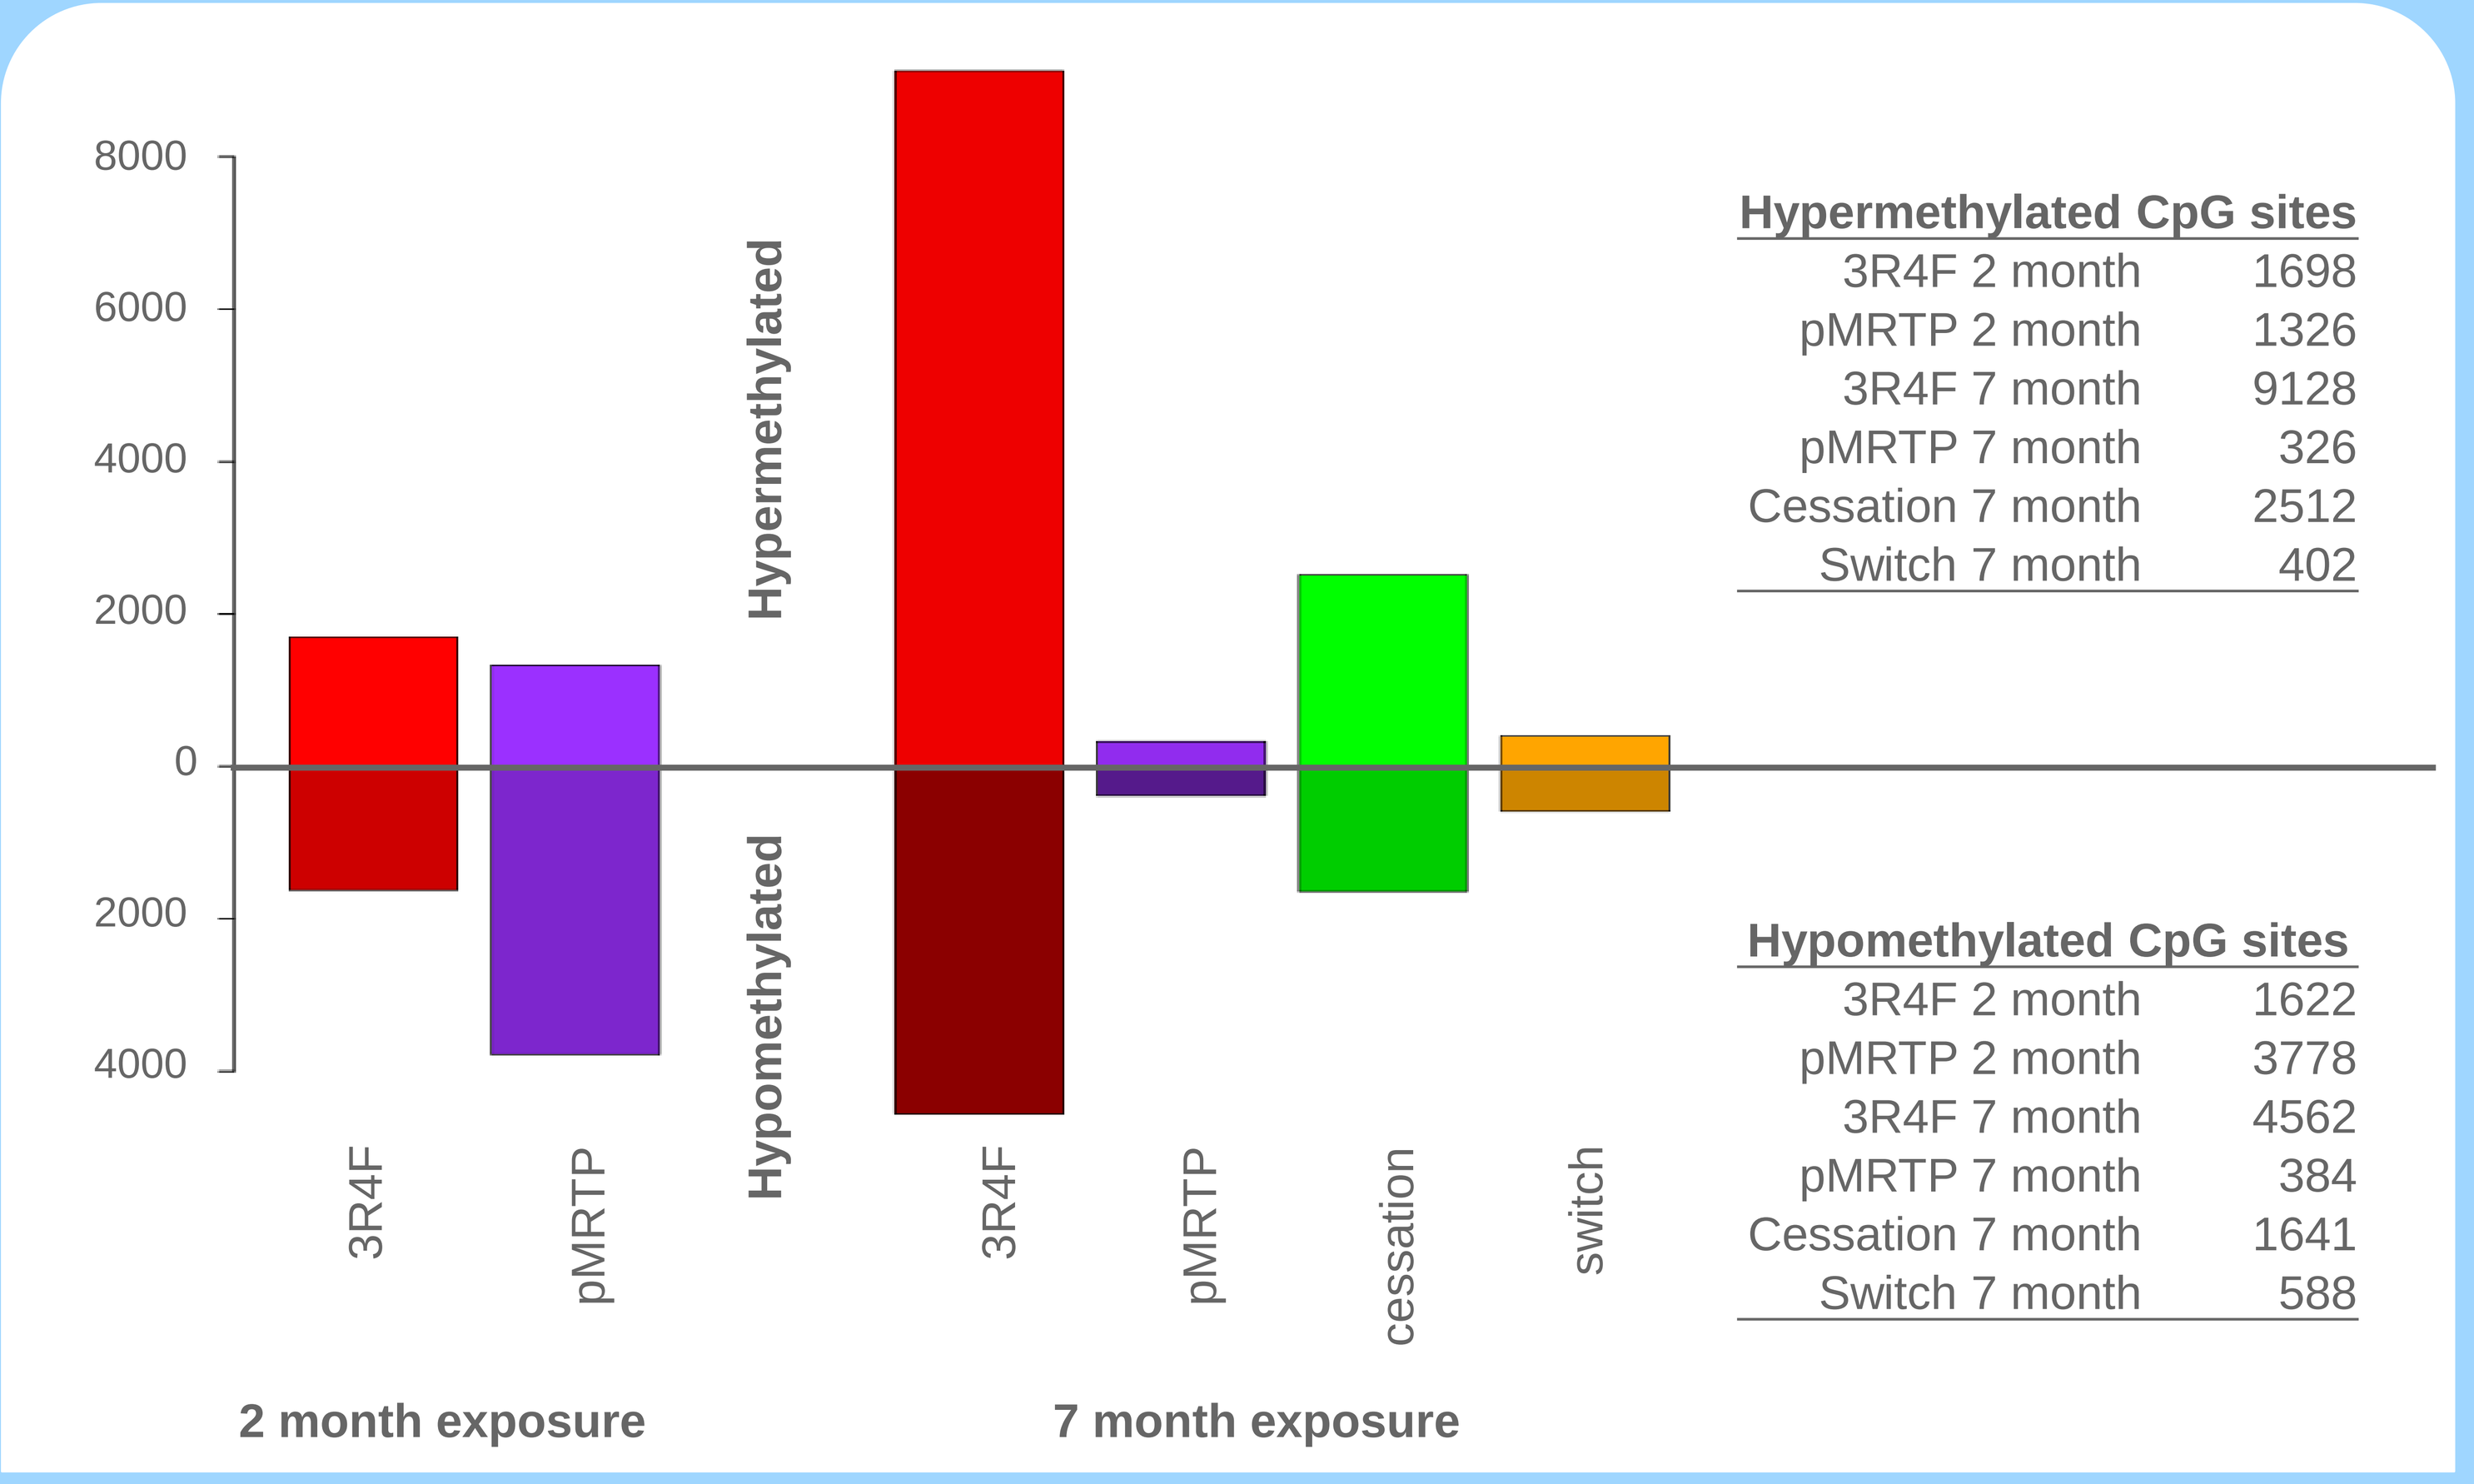
Hypermethylated
Hypomethylated
Hypermethylated CpG sites
3R4F 2 month 1698
pMRTP 2 month 1326
3R4F 7 month 9128
pMRTP 7 month 326
Cessation 7 month 2512
Switch 7 month 402
Hypomethylated CpG sites
3R4F 2 month 1622
pMRTP 2 month 3778
3R4F 7 month 4562
pMRTP 7 month 384
Cessation 7 month 1641
Switch 7 month 588
2 month exposure
7 month exposure

Figure 4: Hyper- and hypo-methylated CpG sites.
Hyper- and hypomethylated sites were identified using RADmeth[12] with a false discovery rate of 0.05.

Although DNA methylation of CpG sites inside CpG islands is known to modulate gene expression, they represent only 5% of all CpG sites. Comparing the location of differentially methylated CpG sites with that of differentially expressed genes did therefore not show a correlation (figure 5). It however showed that after 7 months of exposure, the 3R4F aerosol induced more perturbations than the pMRTP aerosol in the lung tissues of C57BL/6 mice.

Figure 5: Differential methylation and differential expression on the mouse chromosome 1.
Differential gene expression from RNA-seq was obtained using the DESeq2 R package[13]. Locations of down- and up-regulated genes are given in yellow and green respectively, and locations of hypo- and hyper-methylated CpG sites are given in red and blue respectively.

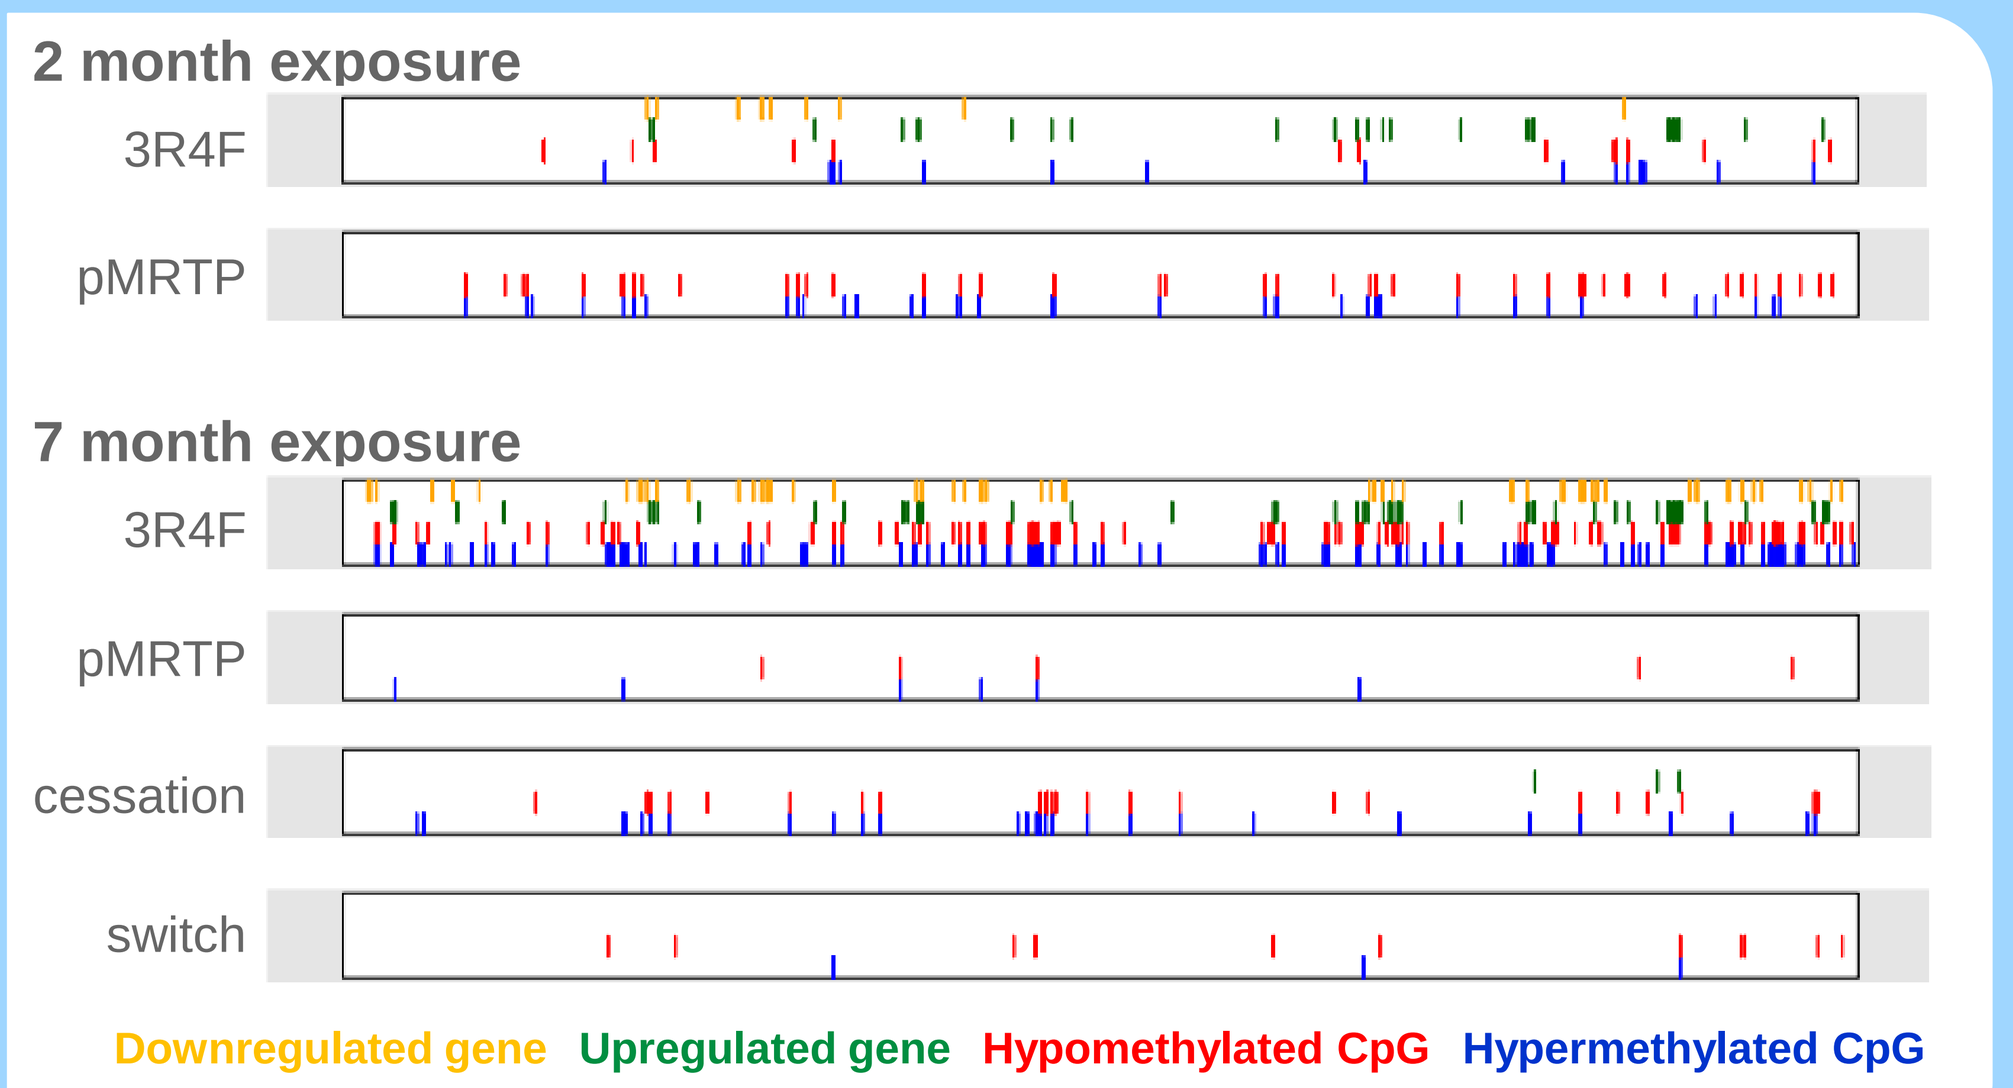
2 month exposure
3R4F
pMRTP
7 month exposure
3R4F
pMRTP
cessation
switch
Downregulated gene
Upregulated gene
Hypomethylated CpG
Hypermethylated CpG

Conclusion

The lungs of mice continuously exposed to aerosol from a reference cigarette presented a larger increase in the amount of hypermethylated CpG sites over time than after either smoking cessation or switching to a pMRTP. The results indicated that smoking cessation or switching to a pMRTP reduced the 3R4F-induced DNA methylation changes in the C57BL/6 mouse lung, and that switching to a pMRTP was closer to smoking cessation than to continuous exposure to 3R4F.

PMI RESEARCH & DEVELOPMENT

SOT2015
San Diego, USA
March 2015

Philip Morris International Research & Development, Quai Jeanrenaud 5, 2000 Neuchâtel, Switzerland
T: +41 58 242 21 11, **F**: +41 58 242 28 11, **W**: www.philipmorrisinternational.com