Physiologically based Pharmacokinetic Modeling of Nicotine from Inhaled Aqueous Aerosol Predicts Marked Gastrointestinal Absorption in Rats

Aditya Reddy Kolli[1*], Florian Martin[1], Yang Xiang[1], Bjoern Titz[1], Ee Tsin Wong[1], Walter K. Schlage[1,2], Emilija Veljkovic[1], Patrick Vanscheeuwijck[1], Julia Hoeng[1*]

[1] PMI Research & Development, Quai Jeanrenaud 5, CH-2000 Neuchâtel, Switzerland

[2] Biology consultant, Max-Baermann-Str. 21, D-51429 Bergisch Gladbach, Germany

Introduction and Objectives

Pharmacokinetics (PK) of compounds from an inhaled aerosol depends on the aerosol deposition fraction as well as on the extent and rate of absorption of a specific compound in the pulmonary region and in the aerodigestive tract and its systemic distribution and clearance. Several factors such as aerosol physiochemical properties, lung morphology and inhalation topography influence aerosol dosimetry. Our objective was to develop and validate a PBPK model that describes the absorption and disposition characteristics of nicotine from inhaled aerosols in rats.

Methods

Aerosol Generation: Nicotine dissolved in PBS was aerosolized using a Collison nebulizer. Aerosol from 3R4F reference cigarettes were generated in a SM2000 (30-port carousel smoking machine) according to the Health Canada Intense Regimen. The target nicotine concentrations of 23 and 50 µg/L were achieved by diluting output of the aerosol generation device with air.

Animal Exposure: As per OECD guidelines for subacute inhalation toxicity testing, 12 male Sprague-Dawley rats per group were exposed (nose-only) to cigarette smoke and aerosols containing 23 and 50 µg/L concentrations of nicotine for 6 hours/day for 5 days/week for 2 weeks.

Measurements: The aerosol particle sizes were determined by using cascade impactors. The plasma concentrations of nicotine and cotinine were monitored 16 hours post-exposure on days 4 and 11.

PBPK model: We built a semi-descriptive rat inhalation PBPK model for inhaled aerosol in R using the *mrgsolve* package [1] and parameters from Plowchalk et.al [2]. The fraction of aerosol (f_0) entering lung and GI, absorption rate from GI (kGI), nicotine metabolism (Vmax) and cotinine elimination (kEL) were fitted using the global optimization package *GenSA* [3] to plasma concentrations. The aerosol deposited fractions were calculated using the MPPD model [4] and compared to PBPK model predictions.

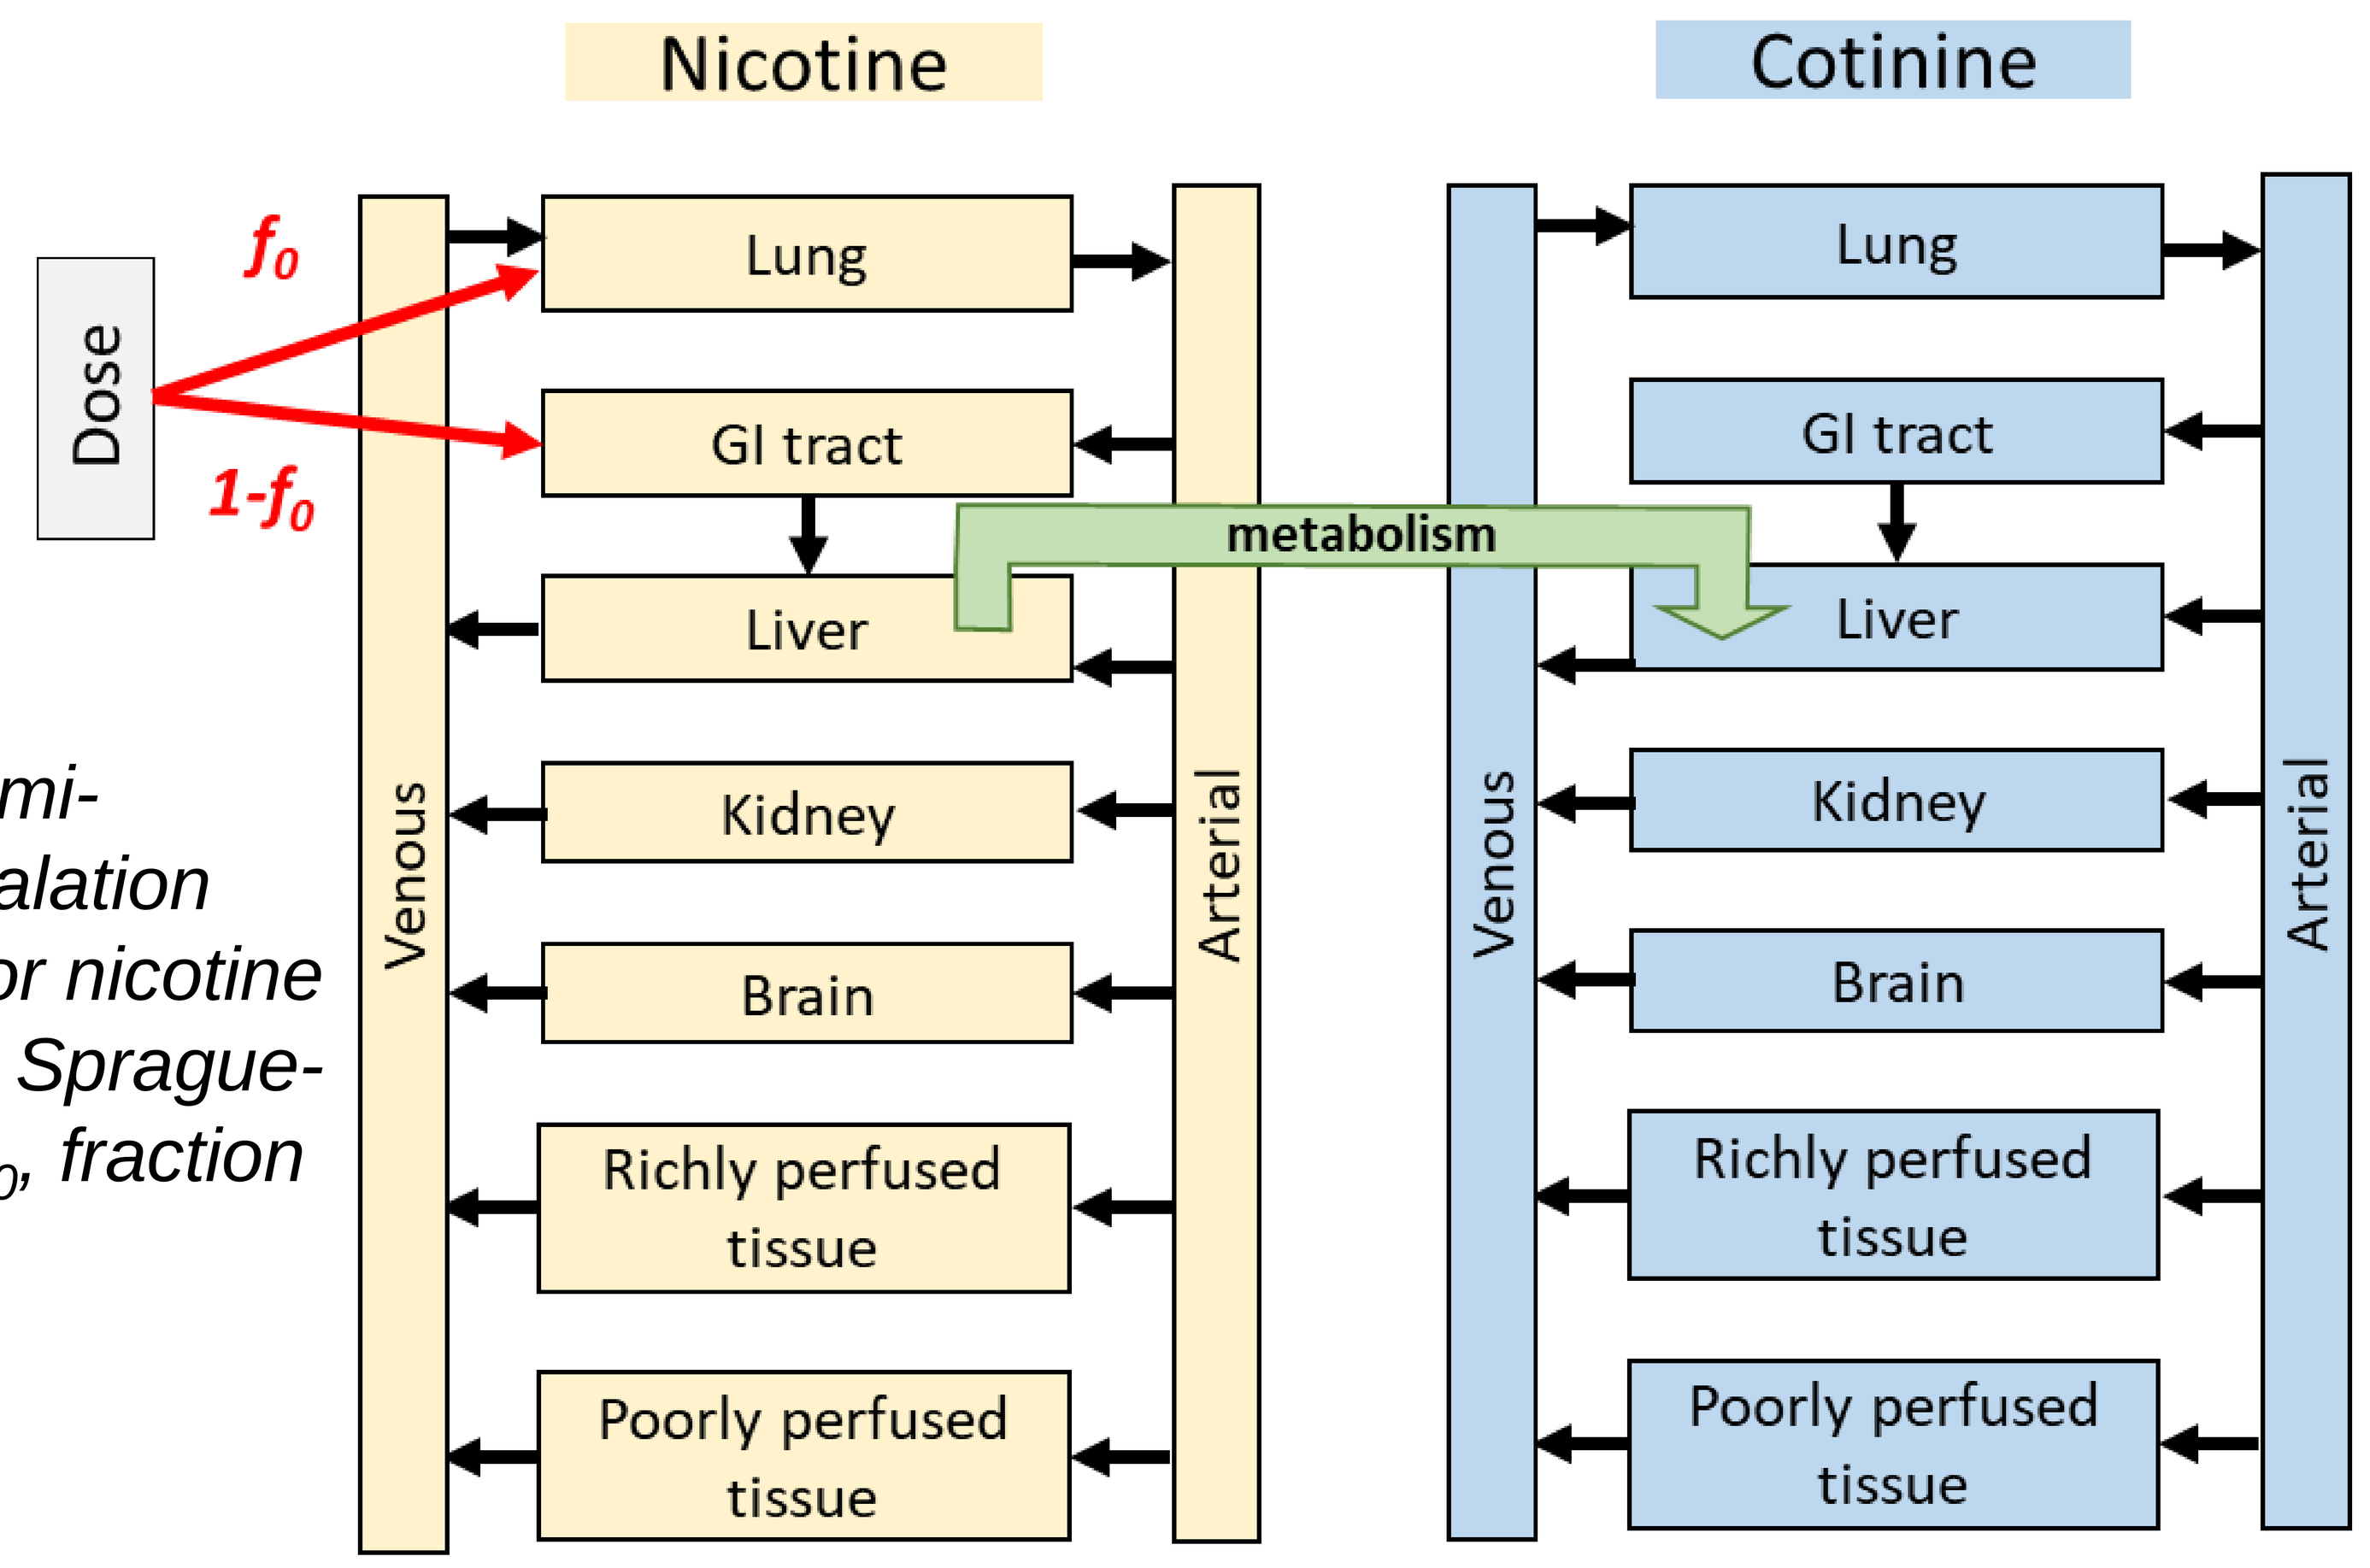
Nicotine
Cotinine
Dose
f_0
$1-f_0$
Venous
Arterial
Lung
GI tract
Liver
Kidney
Brain
Richly perfused tissue
Poorly perfused tissue
metabolism

Figure 1: *A semi-descriptive inhalation PBPK model for nicotine and cotinine in Sprague-Dawley rats. F_0, fraction entering lung*

Results

The simulated plasma concentration-time profiles were in close agreement with experimentally measured data across multiple exposures. Plasma time courses of nicotine and cotinine showed no difference to initial values (Day 4) after 2 weeks of dosing and fell within the model predicted 95% confidence interval. Nicotine C_{max} were 602.9 and 1361.1 ng/mL for 23 and 50 µg/L exposures respectively.

The difference in C_{max} for cigarette smoke and aerosol, despite having same concentrations of nicotine, is hypothesized to arise from ventilation rate changes. The respiratory minute volume (RMV) for rats exposed to smoke were significantly lower (irritancy effect) compared to those exposed to nebulized aerosol. RMV values were accounted for in the model.

PK of Nicotine and Cotinine after Cigarette Smoke Exposure

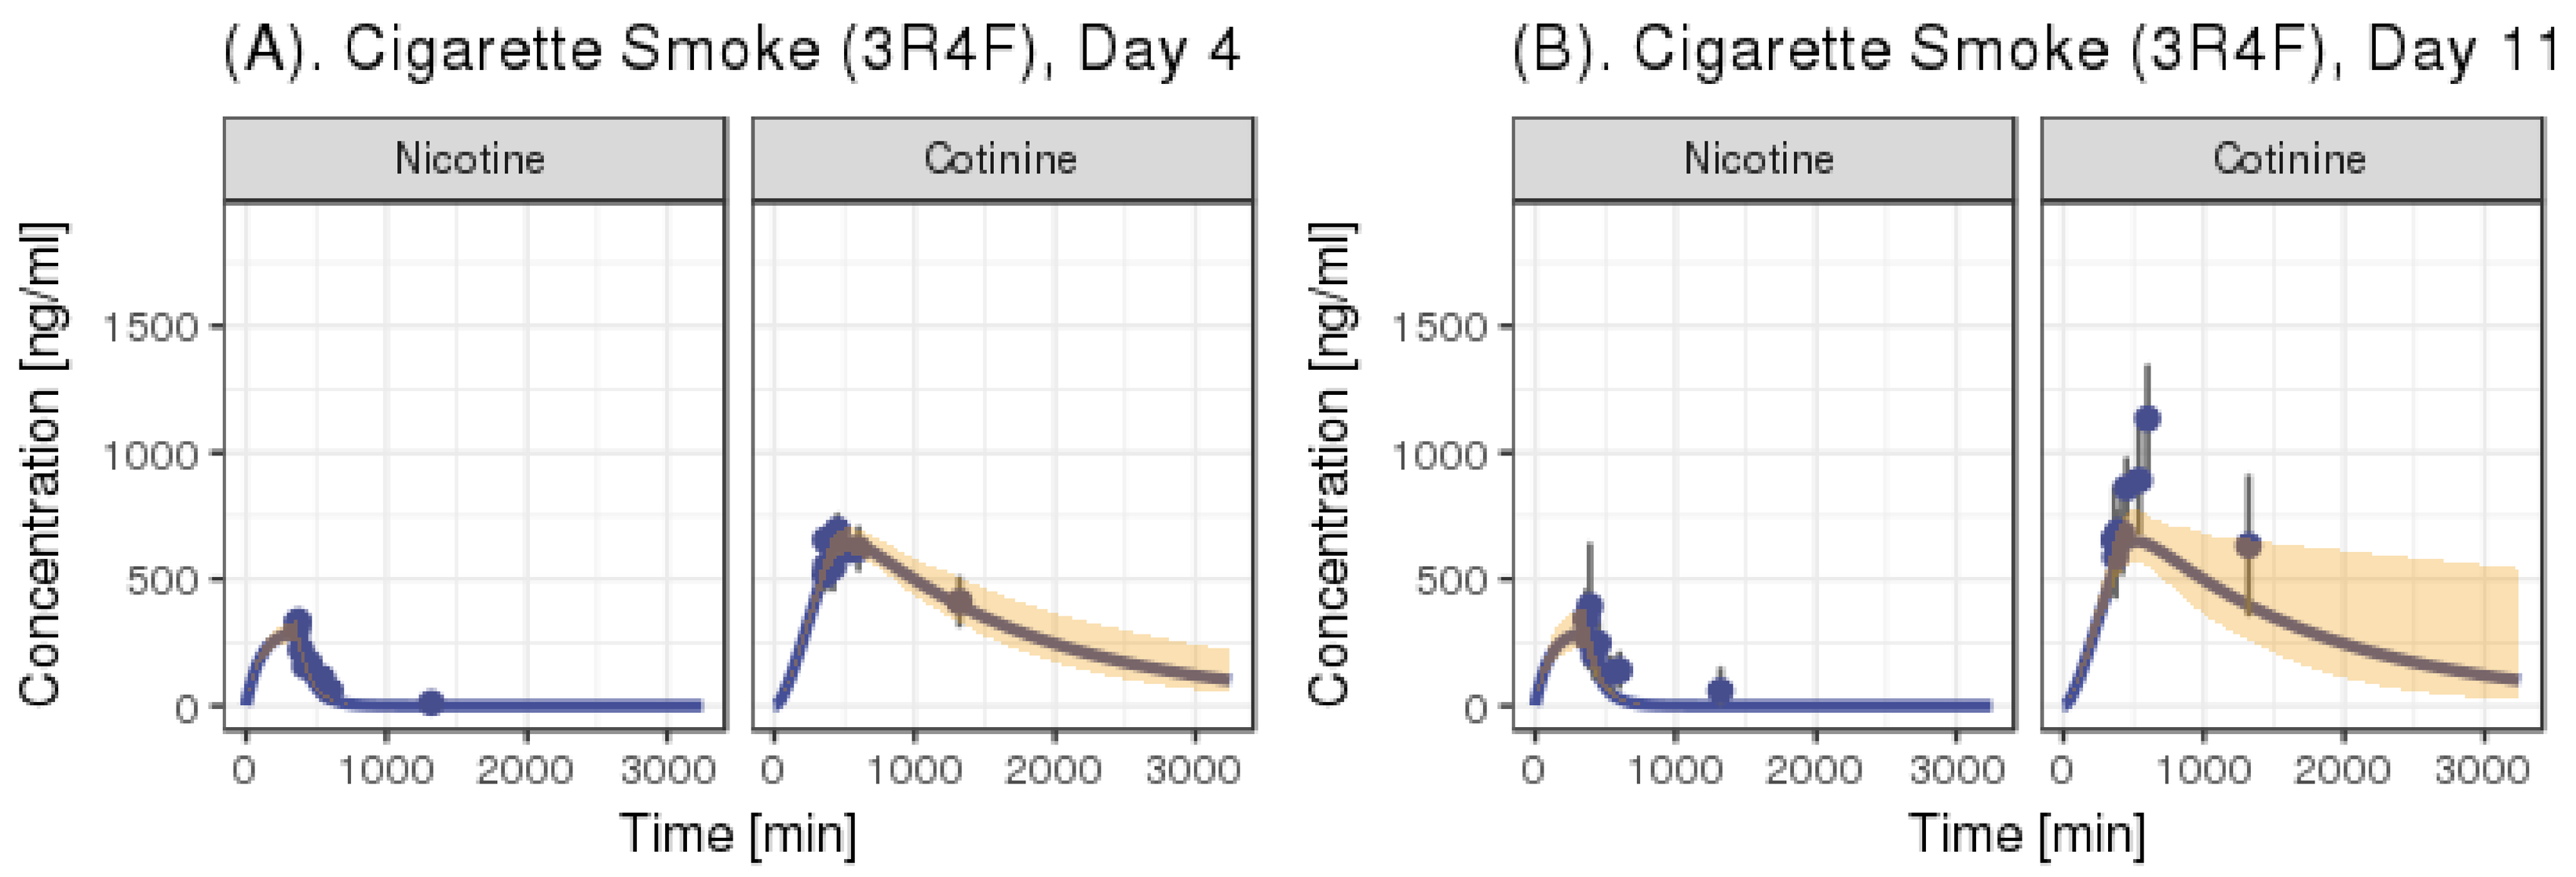
(A). Cigarette Smoke (3R4F), Day 4
(B). Cigarette Smoke (3R4F), Day 11
Nicotine
Cotinine
Concentration [ng/ml]
Time [min]

Figure 2: *Comparison of simulated (lines) and measured (points) nicotine and cotinine plasma concentrations after cigarette smoke exposures on day 4 and 11. Orange band around the curve represents the 95% confidence interval.*

PK of Nicotine and Cotinine after Aerosol Exposure

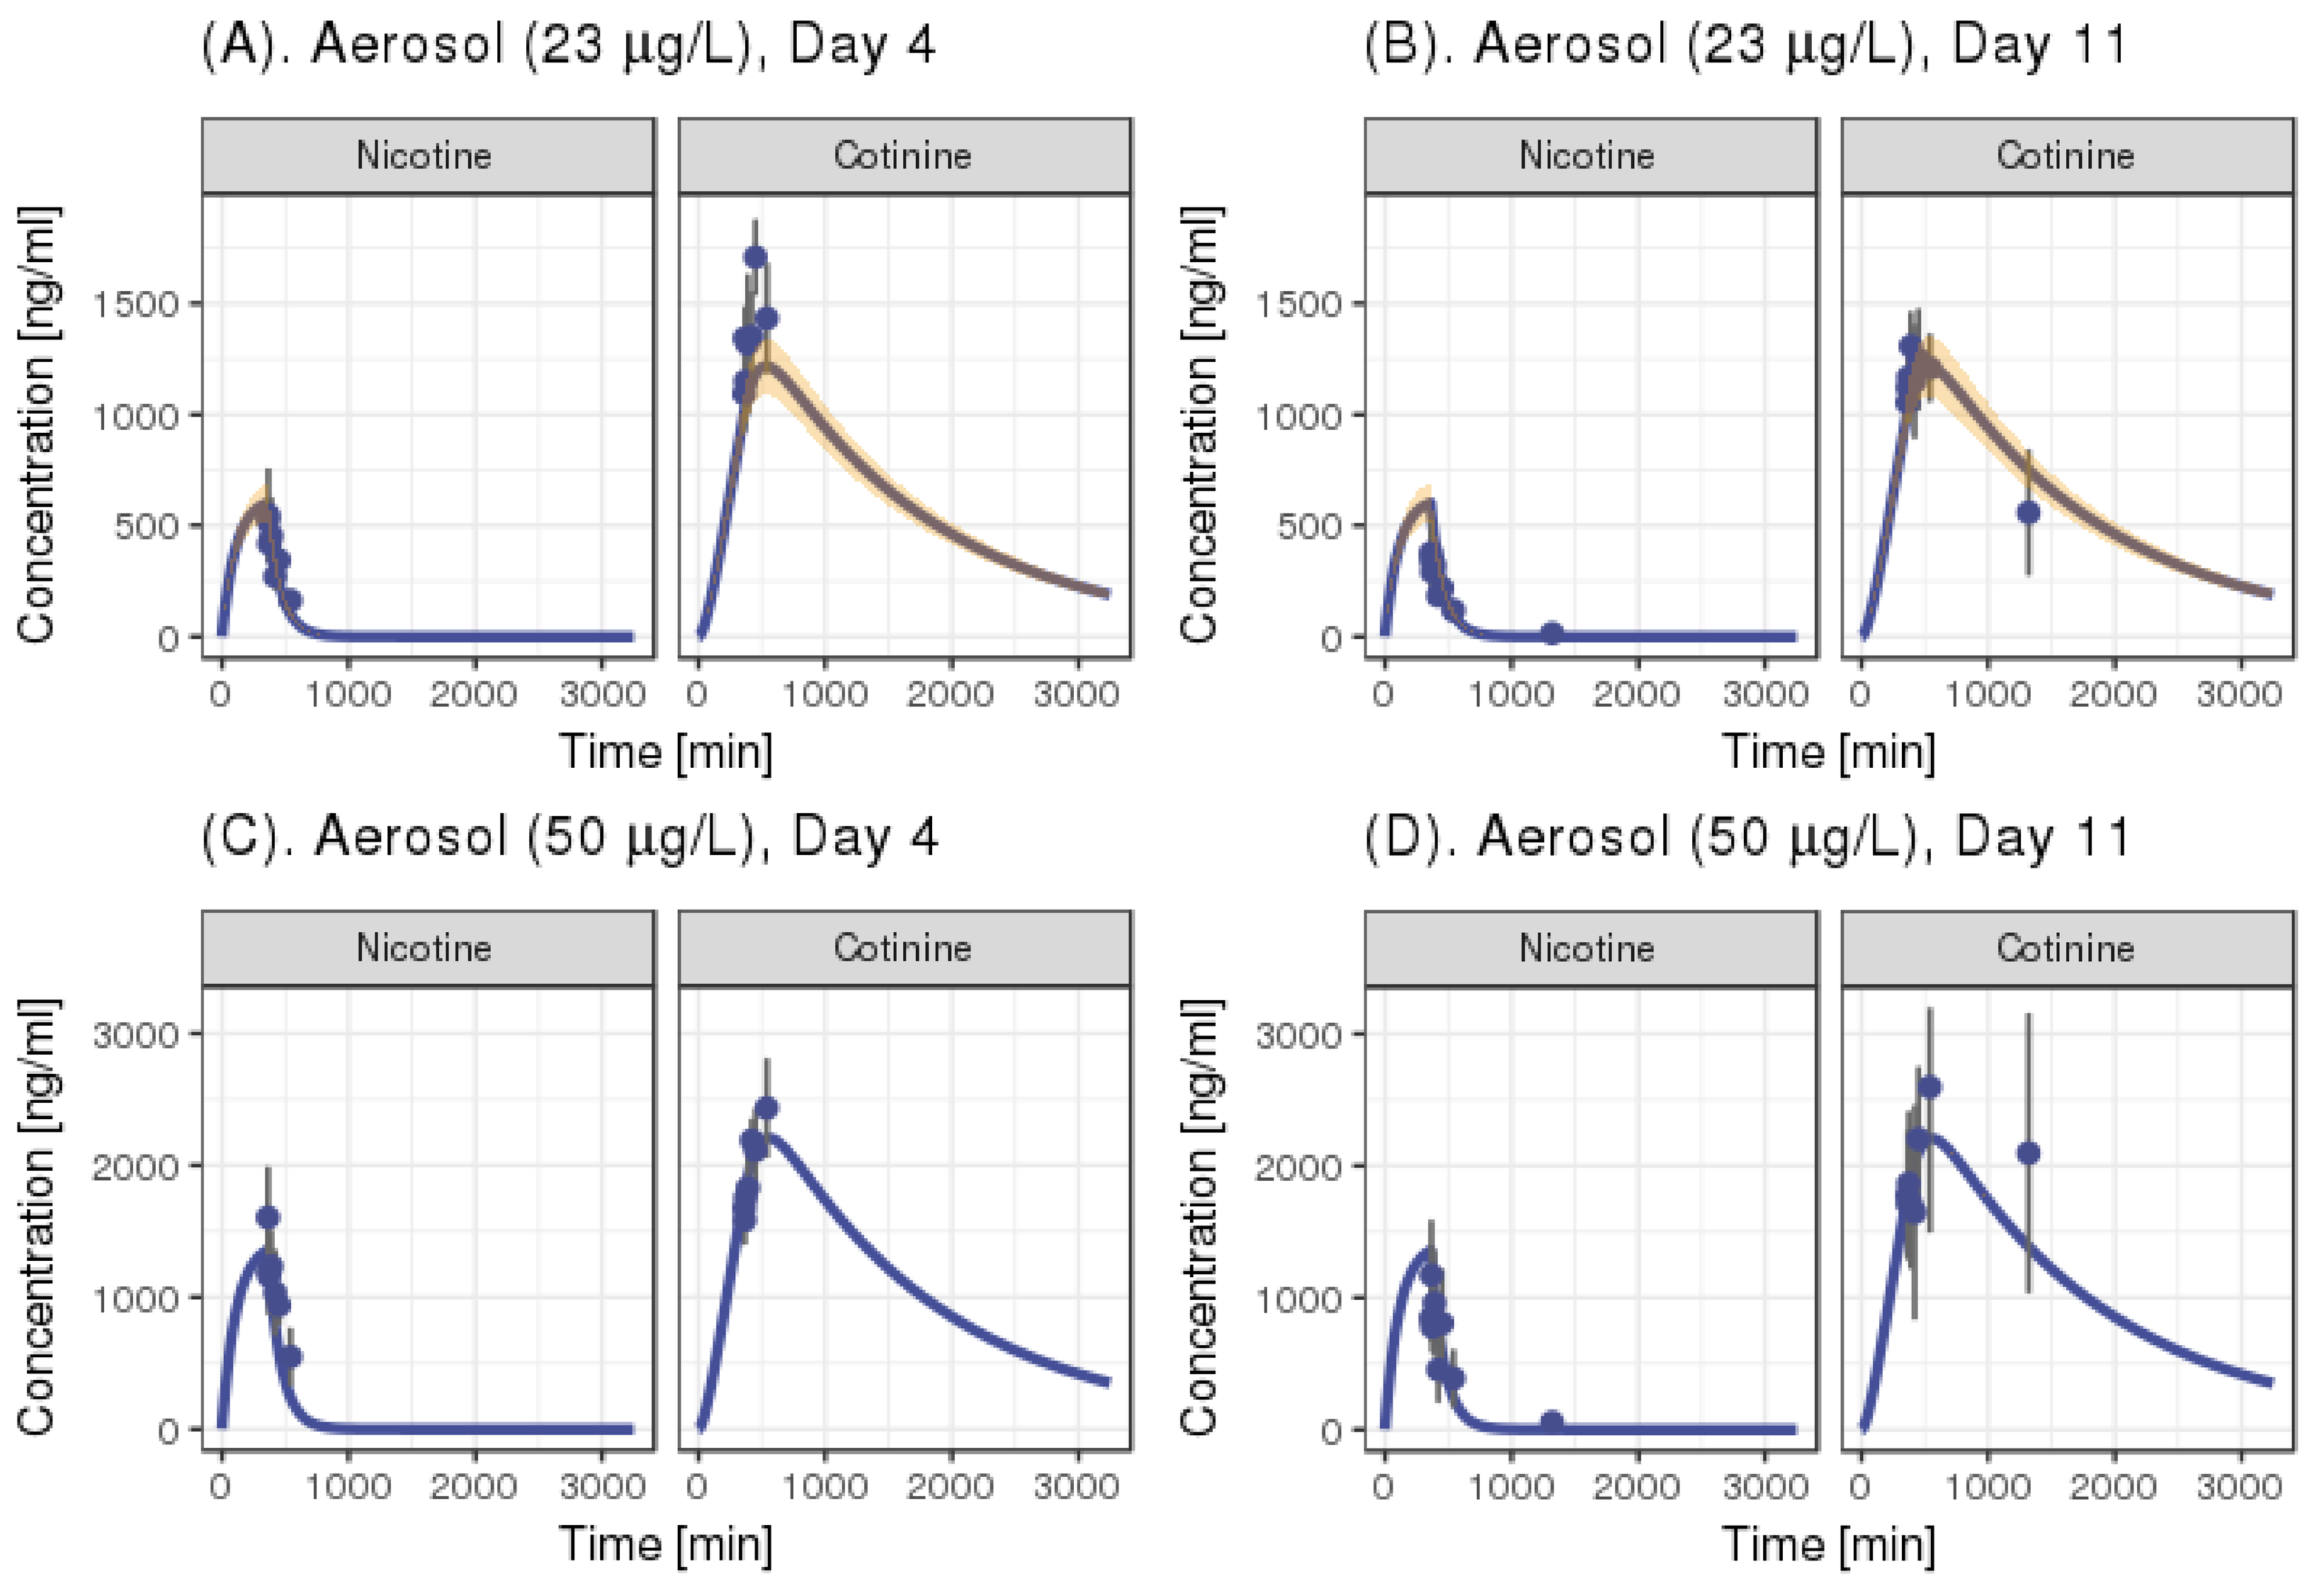
(A). Aerosol (23 µg/L), Day 4
(B). Aerosol (23 µg/L), Day 11
(C). Aerosol (50 µg/L), Day 4
(D). Aerosol (50 µg/L), Day 11
Nicotine
Cotinine
Concentration [ng/ml]
Time [min]

Figure 3: *Comparison of simulated (lines) and measured (points) nicotine and cotinine plasma concentrations after aerosol (nebulized nicotine in water) exposures on day 4 and 11. Orange band around the curve represents the 95% confidence interval.*

Results

The best fit to nicotine and cotinine plasma concentration time courses was obtained by assuming, a 95% deposited and absorption of nicotine in the alveolar space, in line with the literature [5].

Similarly, for nebulized aerosol, 39.6% of nicotine was predicted to be absorbed through gastrointestinal tract at a kGI of 0.175 hr^{-1}. This outcome is inline with additionally performed MPPD model calculations.

The nebulized aerosol had a mean MMAD (microns) of 1.35 ± 0.15 and a mean GSD of 1.91 ± 0.2 while cigarette smoke will have MMAD < 1 (microns) and a GSD of 1.3.

Model Predicted Absorbed/Deposited Fractions of Aerosols

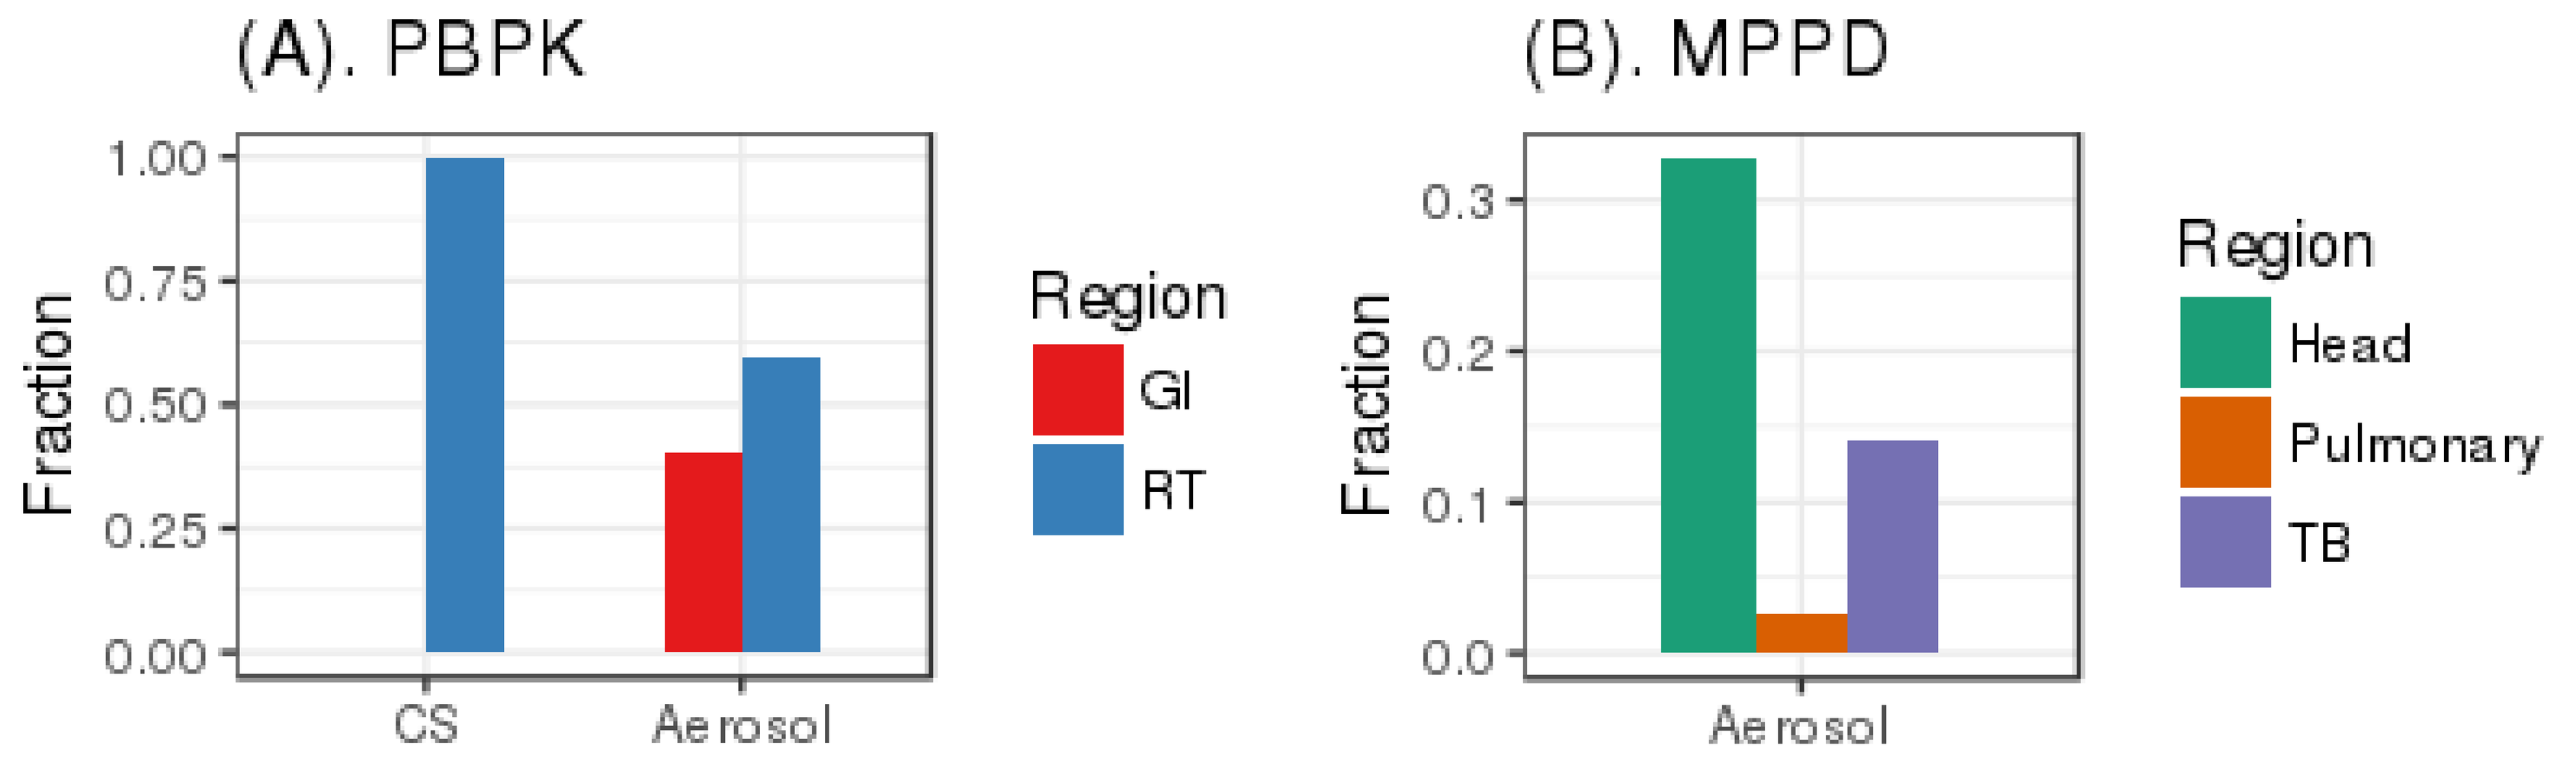
(A). PBPK
(B). MPPD
Fraction
CS
Aerosol
Region
GI
RT
Head
Pulmonary
TB

Figure 4: *PBPK model (A) and MPPD model (B) predicted absorbed and deposited fractions respectively. GI, Gastrointestinal tract; RT, Respiratory tract; CS, Cigarette Smoke; TB, TracheoBronchial;*

Conclusions

- The PBPK model predicted systemic exposures for inhaled aerosols by estimating fractions absorbed in respiratory and gastrointestinal tracts.
- In the PBPK model, partial GI absorption of nicotine from inhaled aqueous aerosol could explain the experimental PK of nicotine/cotinine.
- While the overall agreement of modeled and measured nicotine/cotinine PK was high, further refinements of the model will be tested to elucidate the contribution of the GI tract to the absorption of inhaled nicotine in the rat model

Acknowledgements

We would like to thank Dr. Arkadiusz Kuczaj and Dr. Stefan Lebrun for their project support.

* Corresponding authors Aditya Reddy Kolli (AdityaReddy.Kolli@pmi.com) and Julia Hoeng (Julia.Hoeng@pmi.com)

References

[1] Baron K, mrgsolve 2017.
[2] Plowchalk, DR, Toxicol. Appl. Pharmacol 1992, 116, 177-188.
[3] Xiang Y, R Journal 2013, 5(1), 13-28
[4] MPPD v3.01, Applied Research Associates
[5] Benowitz N, Handb Exp Pharmacol. 2009, (192): 29–60.

Conflicts of interest:

Aditya Reddy Kolli, Florian Martin, Yang Xiang, Bjoern Titz, Ee Tsin Wong, Emilija Veljkovic, Patrick Vanscheeuwijck, Julia Hoeng are all employees of Philip Morris International S.A.
Walter K. Schlage is a consultant for Philip Morris International S.A

American Society for Clinical Pharmacology and Therapeutics, Washington DC | March 13-16, 2019

Competing Financial Interest – The research described in this poster was sponsored by Philip Morris International