Effects of Cigarette Smoke on the High Fat Diet-Induced ApoE-/- Mouse Model of Diabetic Atherosclerosis

Danilal Sharma[2], Anita Iskandar[1], Blaine Phillips[2], Hector De Leon[1], Emilija Veljkovic[1], Maciej Cabanski[1], Julia Hoeng[1], Manuel C. Peitsch[1]
[1] Philip Morris International R&D, Quai Jeanrenaud 5, CH-2000, Neuchâtel, Switzerland
[2] Philip Morris International Research Laboratories, Pte Ltd. 50 Science Park Road, Singapore 117400

Introduction

Type 2 diabetes mellitus (T2DM) is characterized by both insulin resistance (IR) and a compensatory hyperinsulinemia (Noto 2013). Smoking has been associated with reduced insulin sensitivity or increased IR, as well as increased plasma glucose (Cena 2013). Studies have reported that cigarette smoking is a strong independent risk factor for T2DM (Cena 2013). Moreover, both smoking and diabetes are known risk-factors for cardiovascular disease (Qazi and Malik, 2013). Recently, the report of the US Surgeon General (USSG, 2014) indicated that the risk of diabetes increases 30-40% in smokers as compared to nonsmokers. Nevertheless, the mechanisms of cigarette smoke (CS)-induced IR are poorly understood (Bergman 2012). Because atherosclerotic cardiovascular disease is known to be the leading cause of morbidity and mortality among patients with diabetes, the apolipoprotein (Apo)E-/- mice would be more relevant as a model for studying diabetes than mice that do not develop atherosclerosis (Li 2011). The high fat diet (HFD)-fed ApoE-/- mice exhibit characteristics of diabetes and atherosclerosis; therefore, utilization of this mouse model could be relevant to examine the mechanism of CS-induced IR in the context of CVD development. Here, we report the effects of CS exposure for 11 weeks in the HFD-fed ApoE-/- mice. We hypothesized that CS would exacerbate the IR-related changes in the HFD-fed mice.

Materials & Methods

Experimental Design

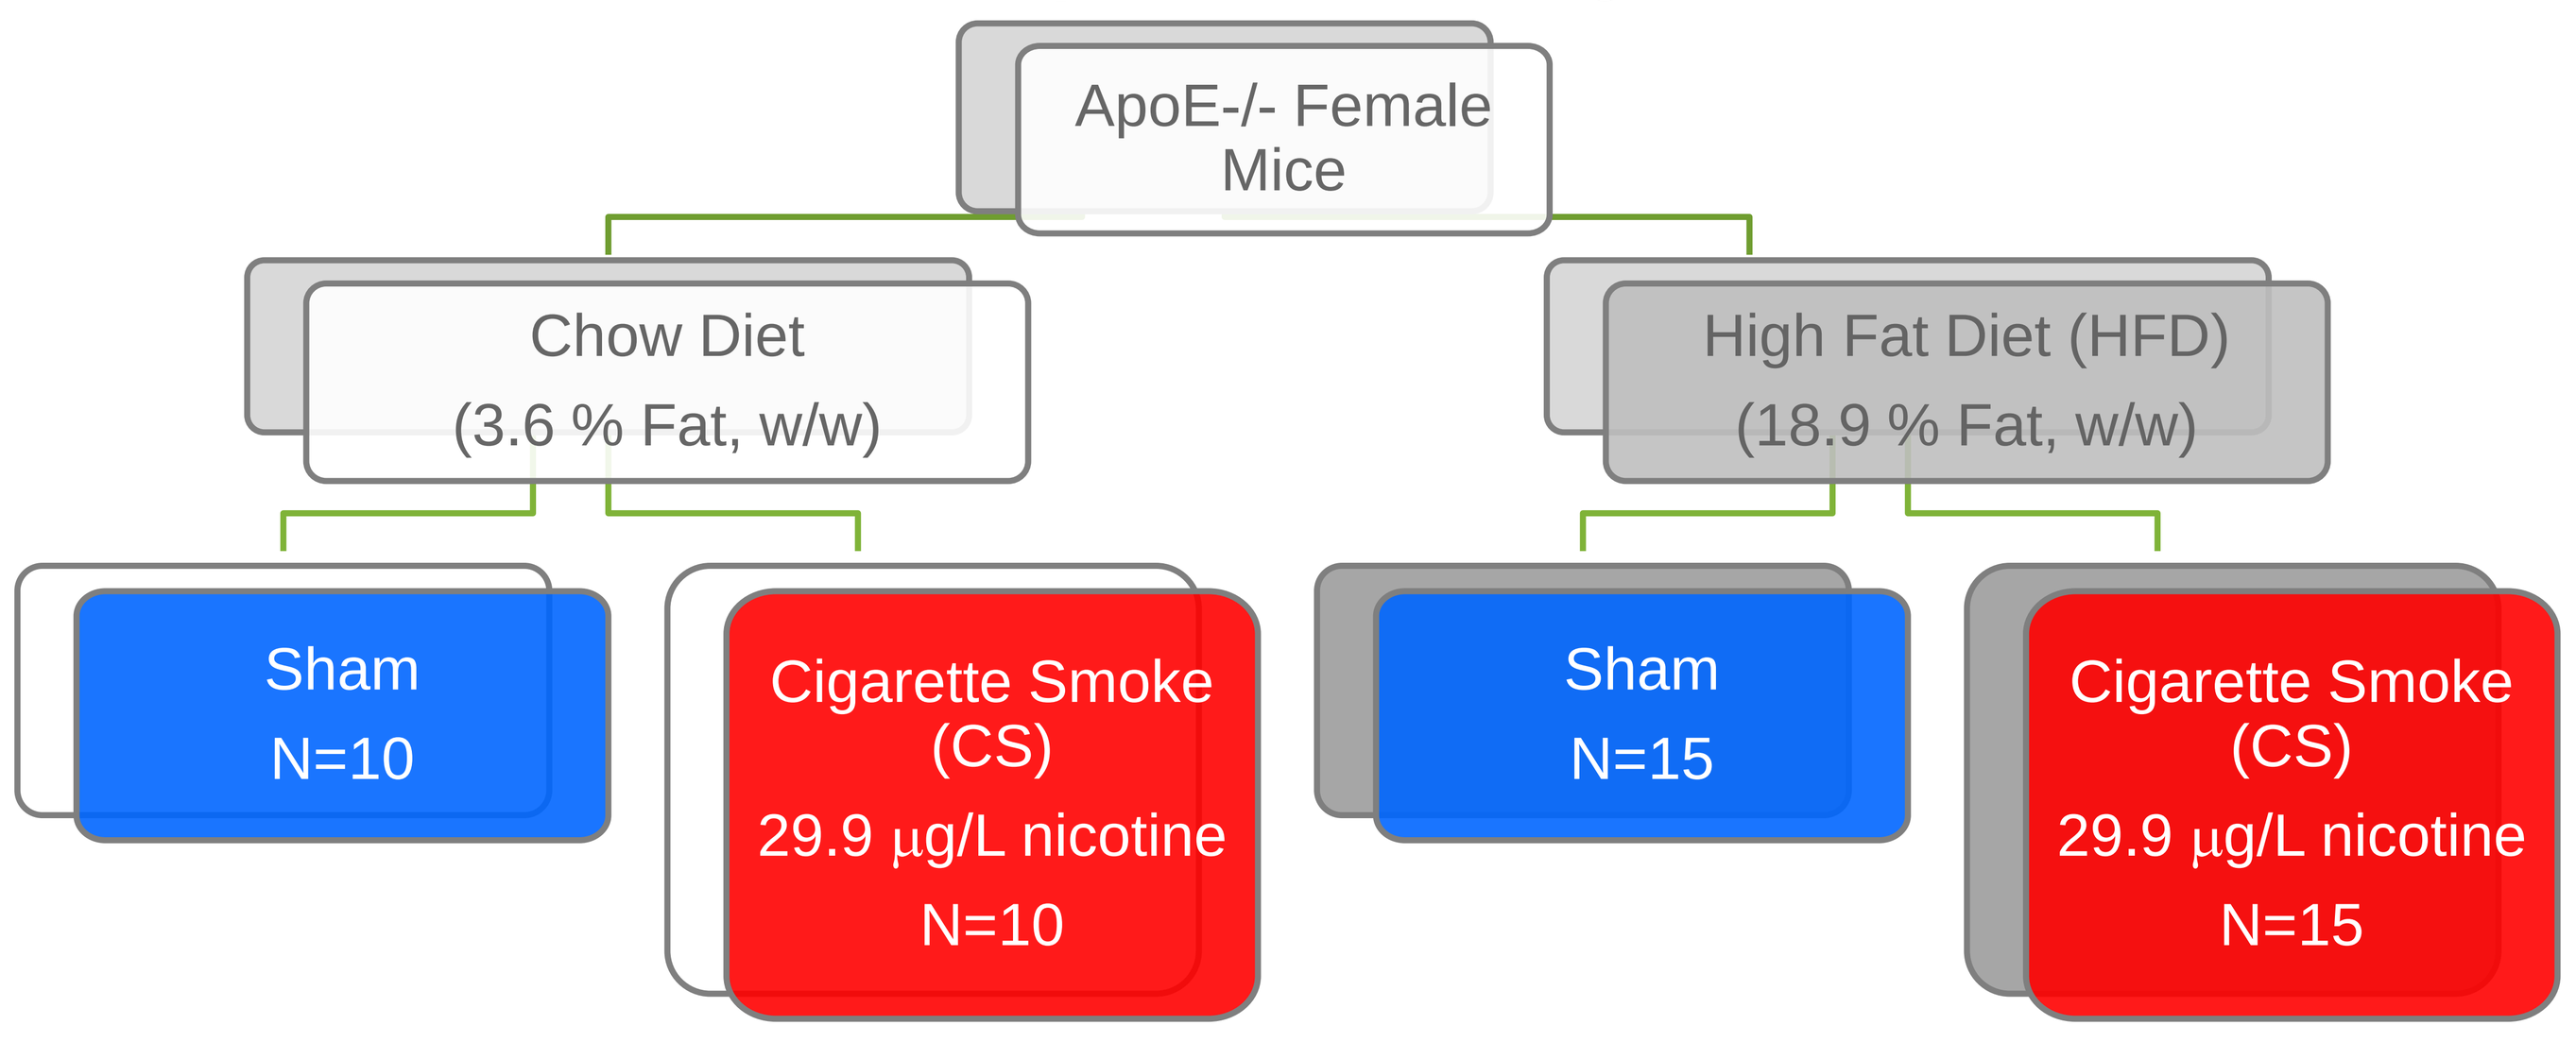
ApoE-/- Female Mice
Chow Diet
(3.6 % Fat, w/w)
High Fat Diet (HFD)
(18.9 % Fat, w/w)
Sham
N=10
Cigarette Smoke (CS)
29.9 µg/L nicotine
N=10
Sham
N=15
Cigarette Smoke (CS)
29.9 µg/L nicotine
N=15

Experimental Procedure

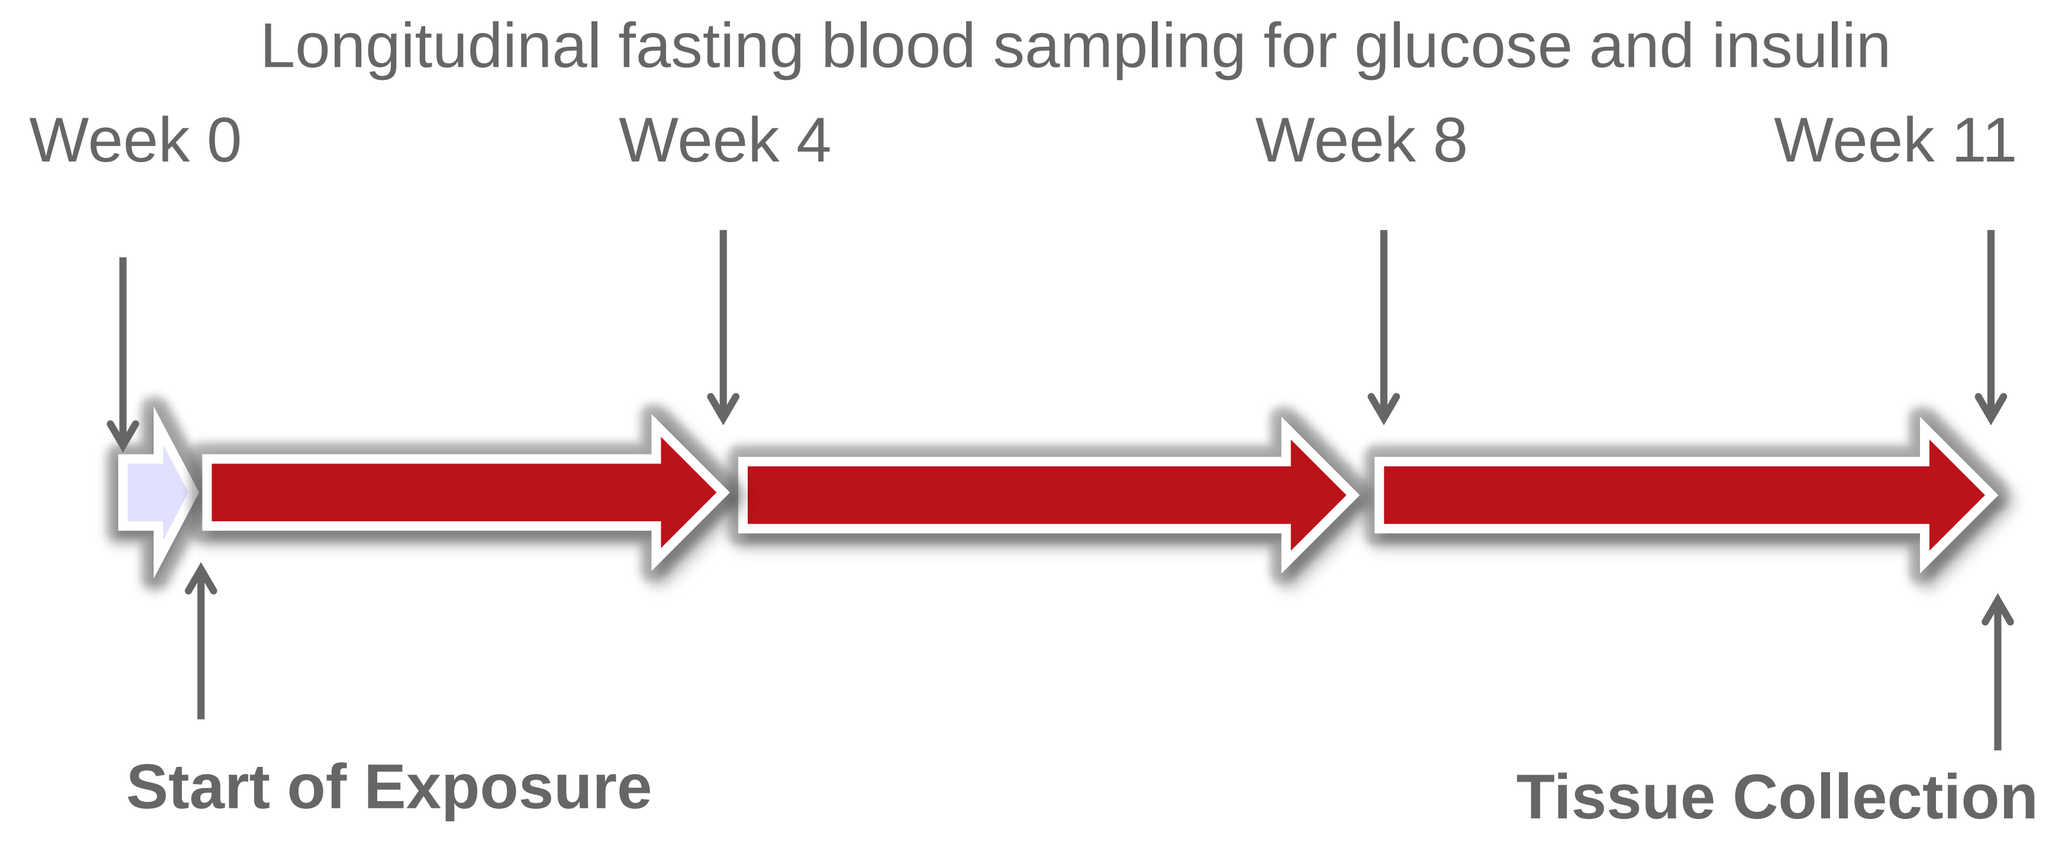
Longitudinal fasting blood sampling for glucose and insulin
Week 0
Week 4
Week 8
Week 11
Start of Exposure
Tissue Collection

Animals – Female B6.apoE-/- mice of 6-8 weeks old were obtained from Taconic Farms USA bred under specific pathogen free conditions. All procedures were carried out in accordance with current guidelines of National Advisory Committee on Laboratory Animal Research (NACLAR) and approved by Phillip Morris International Institutional Animal Care and Use Committee.
Exposure – Animals were exposed to mainstream smoke from 3R4F University of Kentucky reference cigarettes, or exposed to fresh air (sham) for 11 weeks (3 hours daily, 5 days/week, whole body exposure). Diets were removed during the exposure period.
Diets - A pellet T2914C rodent diet was fed to the chow group and a high fat TD.88137 rodent diet to the HFD group (Harland, U.K.).
Glucose – Longitudinal fasting glucose levels were measured in whole blood using a handheld glucometer (Accu Check® Performa, Roche). Mice were fasted for 6 hours before blood collection.
Insulin – Longitudinal fasting insulin levels were measured in plasma using Low Range Assay of the Ultra-Sensitive Mouse Insulin ELISA Kit (Cat # 90080, Crystal Chem, Inc.). Mice were fasted for 6 hours before blood collection.
Lipid Parameters – Cholesterol, triglyceride, HDL, LDL levels were measured at the end of the study in the plasma samples using Beckman Unicell® DXC 600 clinical auto analyzer.
Serum Analytes – Metabolic biomarkers were measured at the end of the study in the serum samples using multiplexed immunoassays.
Aortic Plaque – Plaque size was identified as oil red O-positive material in the aortic arch and morphometric evaluation of total plaque area is done using Visopharm, an image analysis software.
Islet measurement – Islet area measurement was done using Visiopharm image analysis on the H&E stained-slides.

Results

Body Weights

HFD-fed mice had increased body weights as compared with the chow-fed mice. CS exposure reduced the body weights of the chow-fed mice; whereas, CS exposure did not alters the body weights of the HFD-fed mice. Mean ± SD are shown.

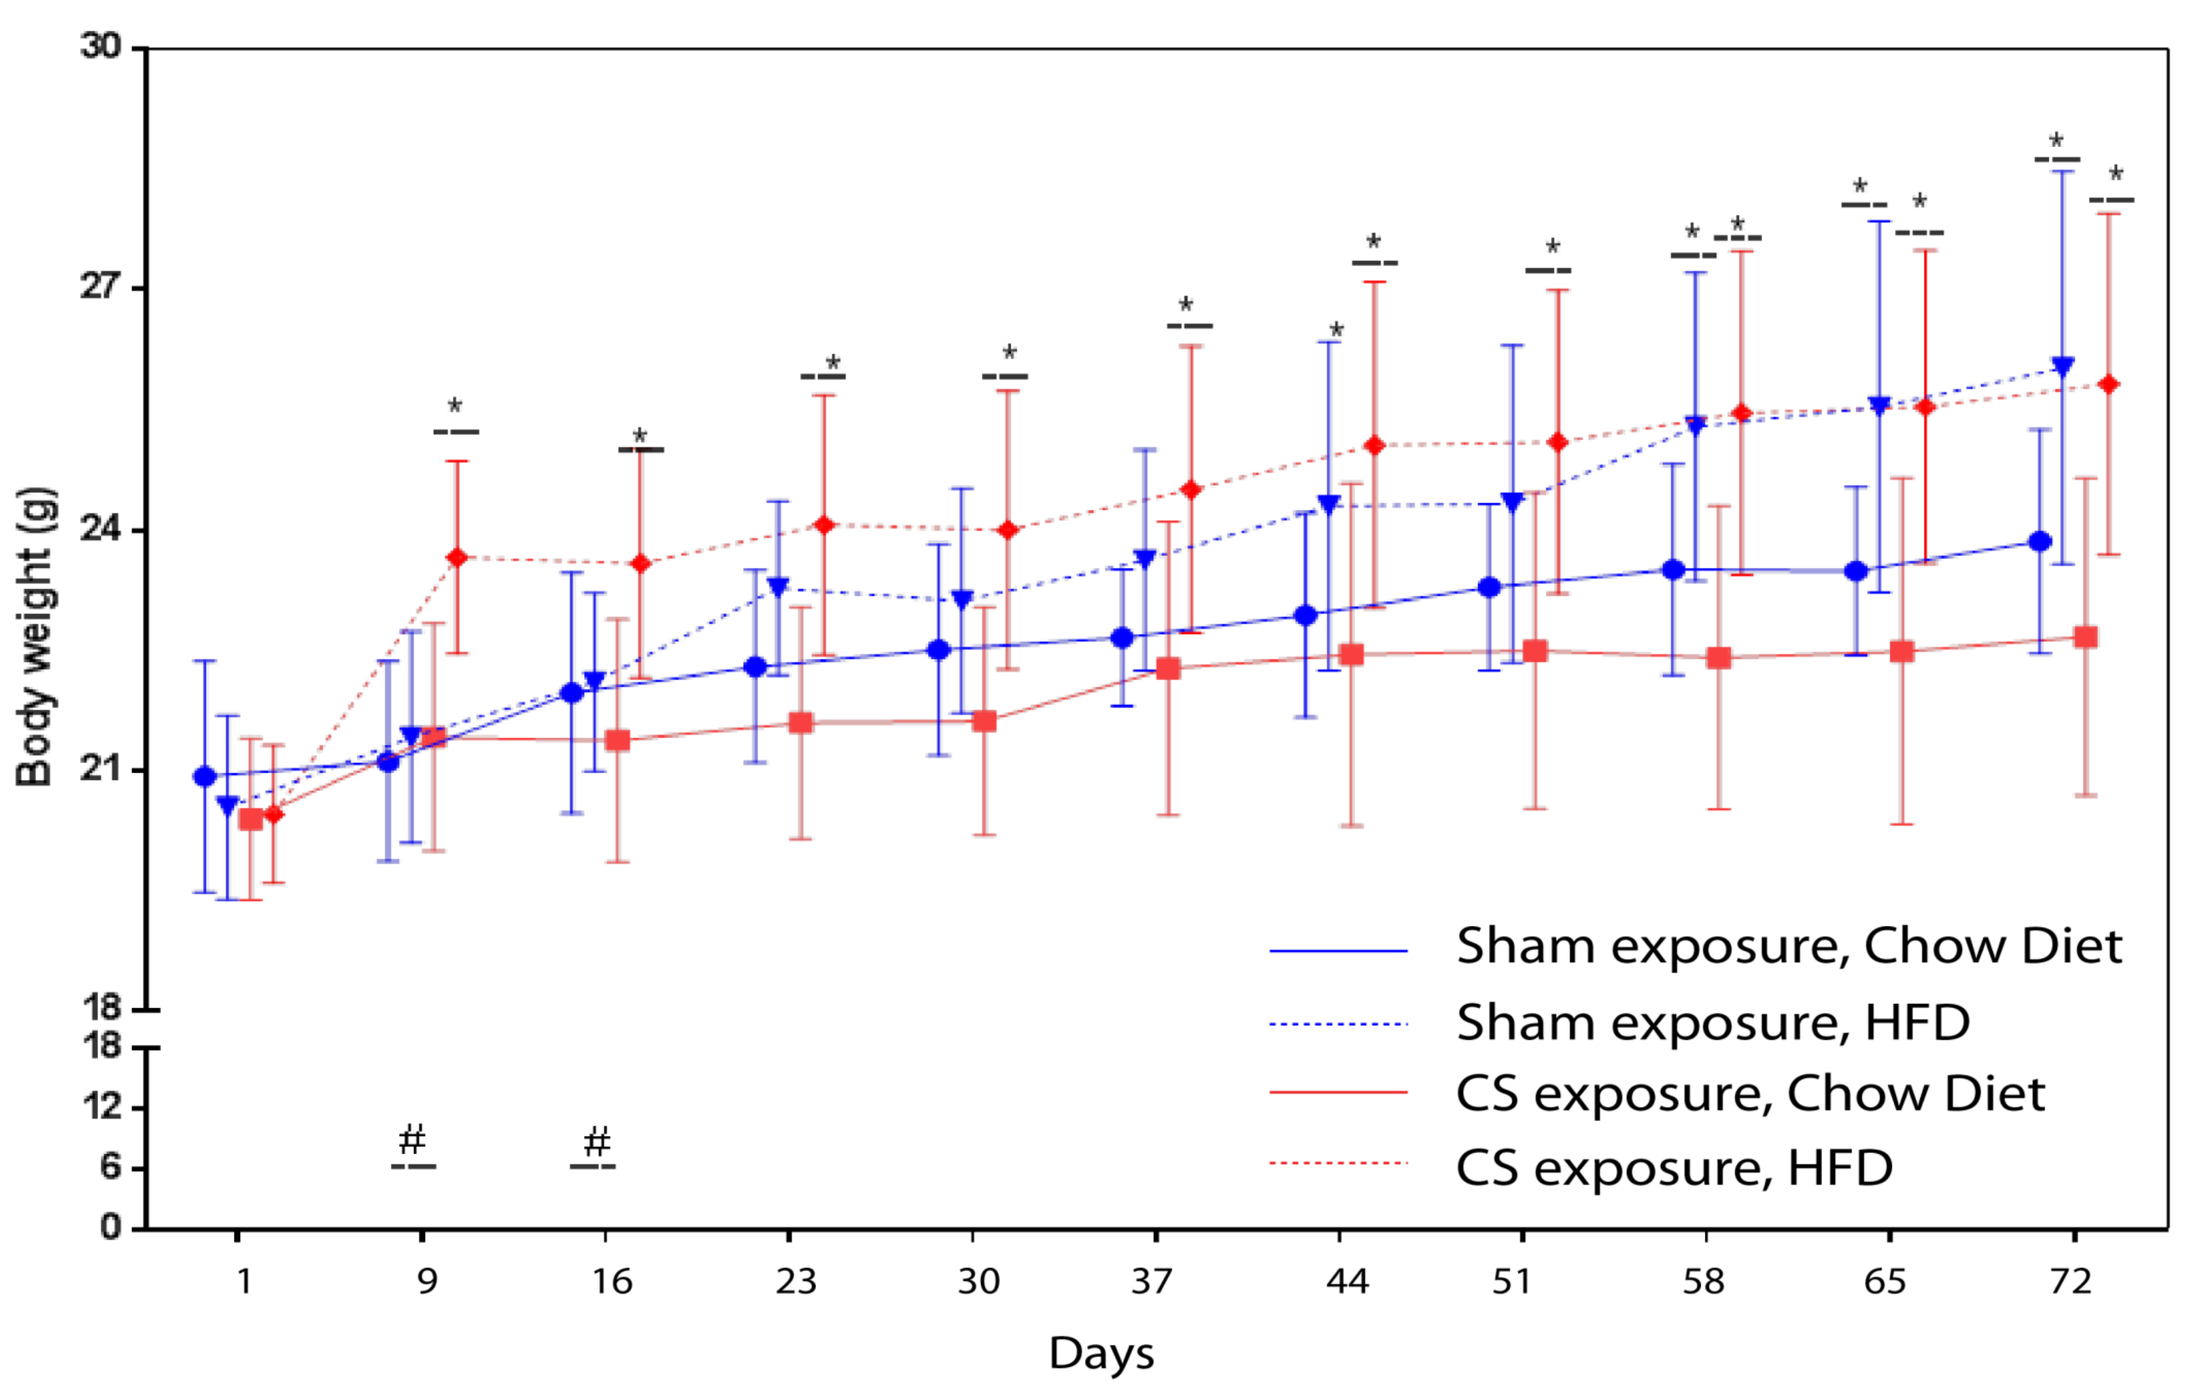
Body weight (g)
Days
Sham exposure, Chow Diet
Sham exposure, HFD
CS exposure, Chow Diet
CS exposure, HFD

* $p < 0.05$ as compared to the respective chow-fed group (for the same exposure).
\# $p < 0.05$ between the CS-exposed HFD and sham-exposed HFD.
Measured by a linear model adjusting for the variability between groups.

Fasting Glucose Levels

At the end of the study, HFD-fed mice had increased fasting glucose levels as compared with the chow-fed mice. CS exposure was associated with reduced fasting glucose levels in both chow- and HFD-fed mice. Mean ± SD are shown.

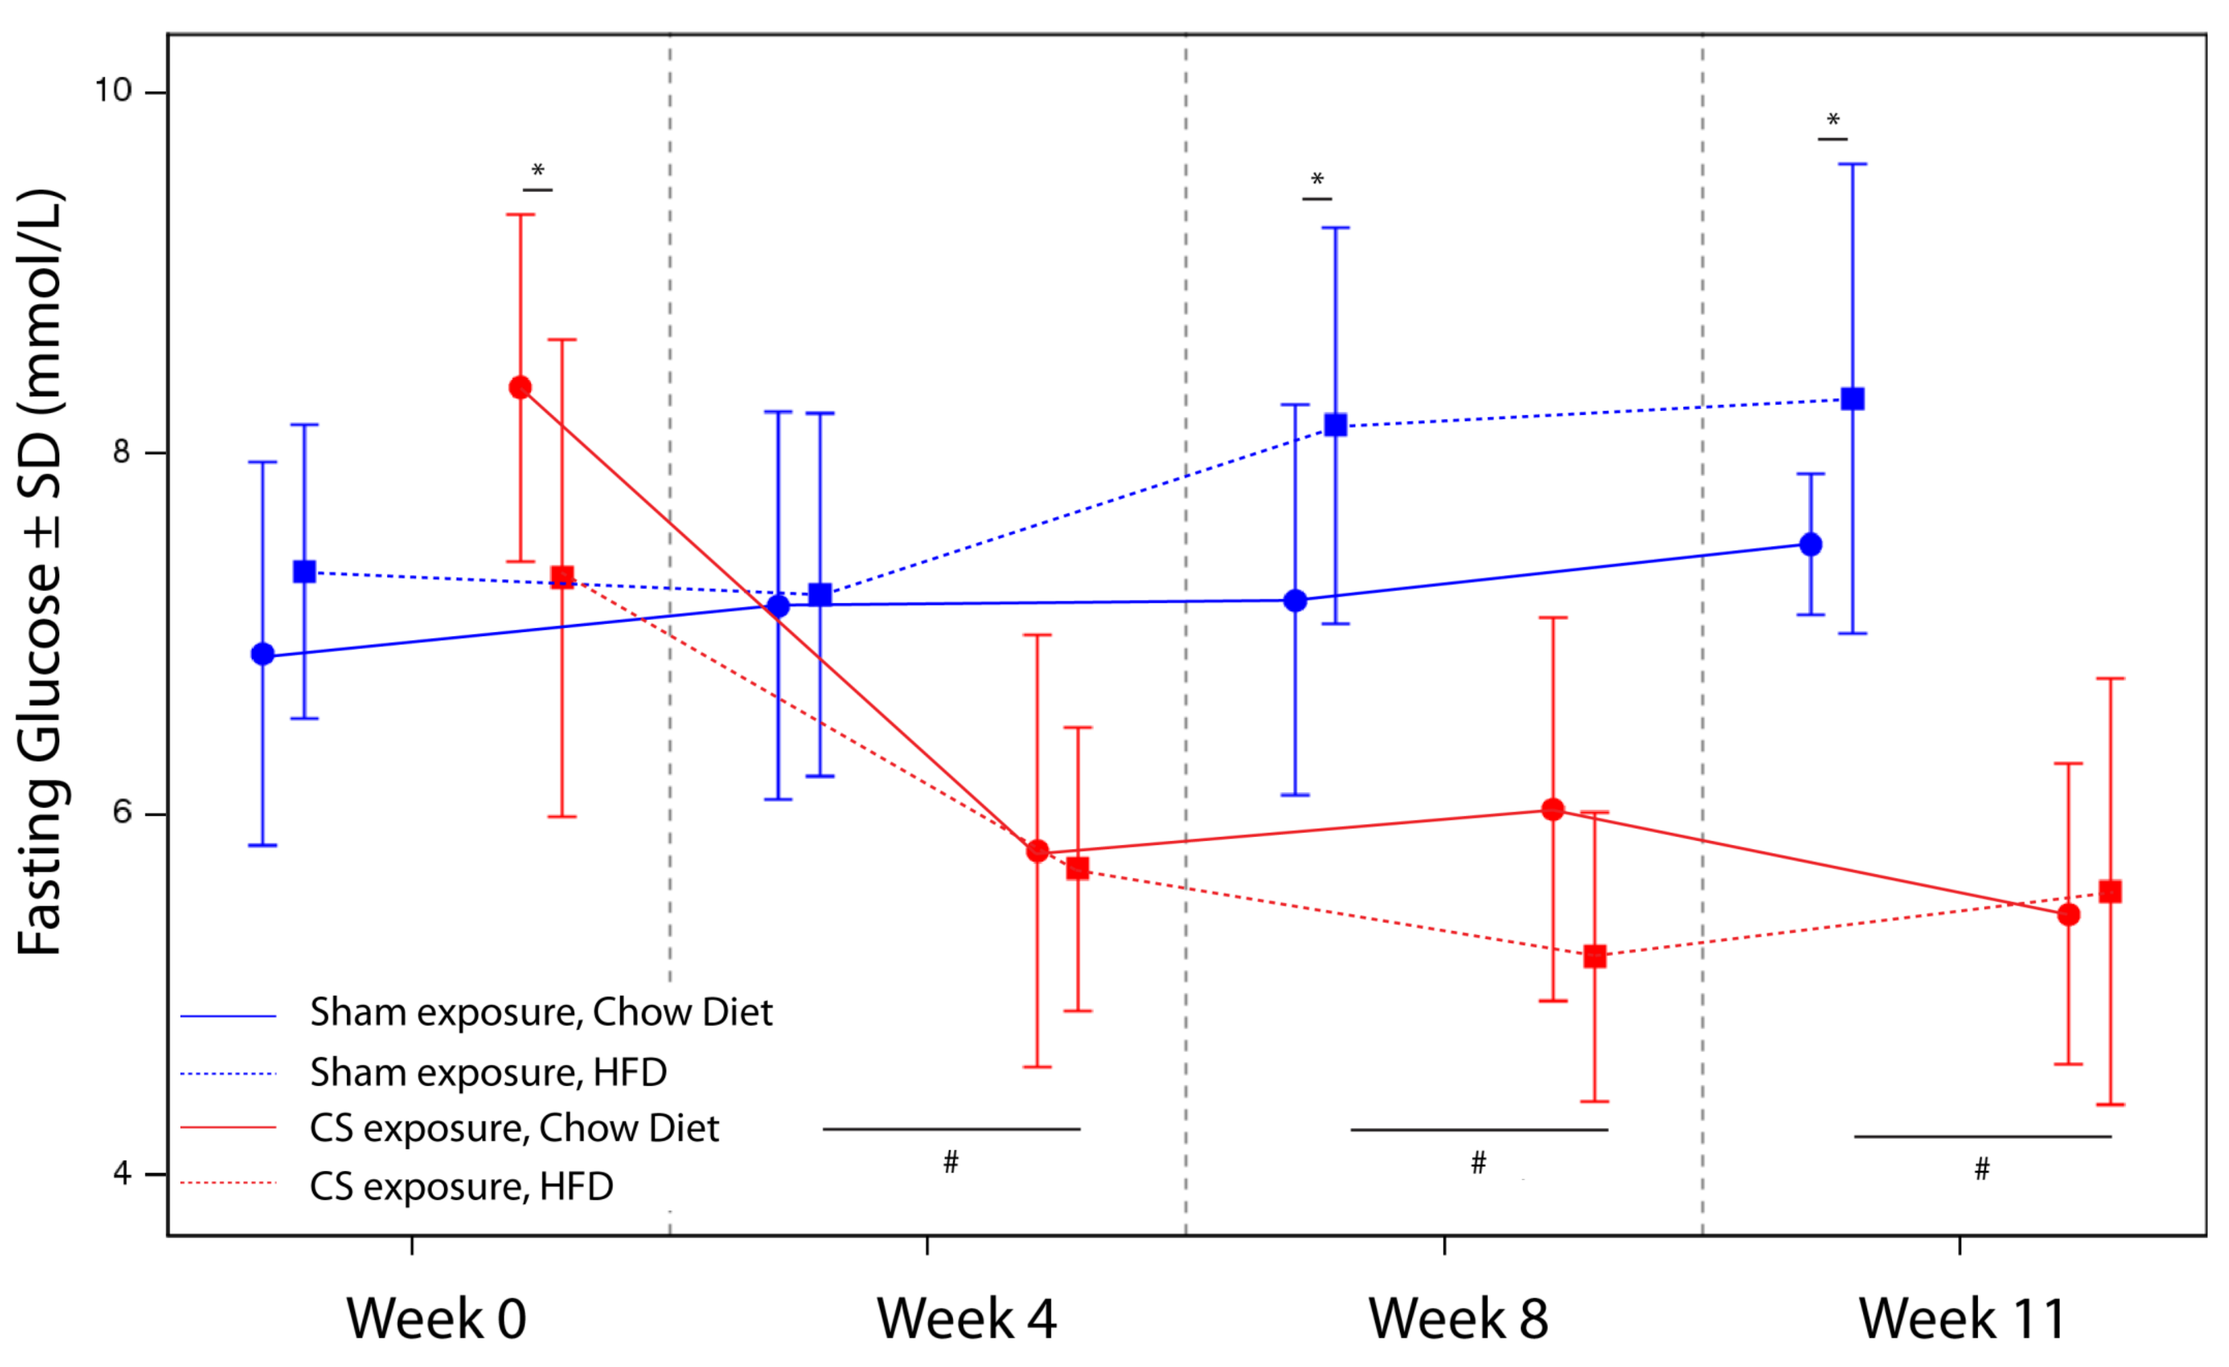
Fasting Glucose ± SD (mmol/L)
Week 0
Week 4
Week 8
Week 11
Sham exposure, Chow Diet
Sham exposure, HFD
CS exposure, Chow Diet
CS exposure, HFD

* $p < 0.05$ as compared to the respective chow-fed group (for the same exposure).
\# $p < 0.05$ between the CS-exposed HFD and sham-exposed HFD.
Measured by a linear model adjusting for the variability between groups.

Fasting Insulin Levels

At the end of the study, HFD-fed mice had increased fasting insulin level as compared with the chow-fed mice. CS exposure was not associated with alterations of fasting insulin levels in both chow-and HFD-fed mice. Mean ± SD are shown.

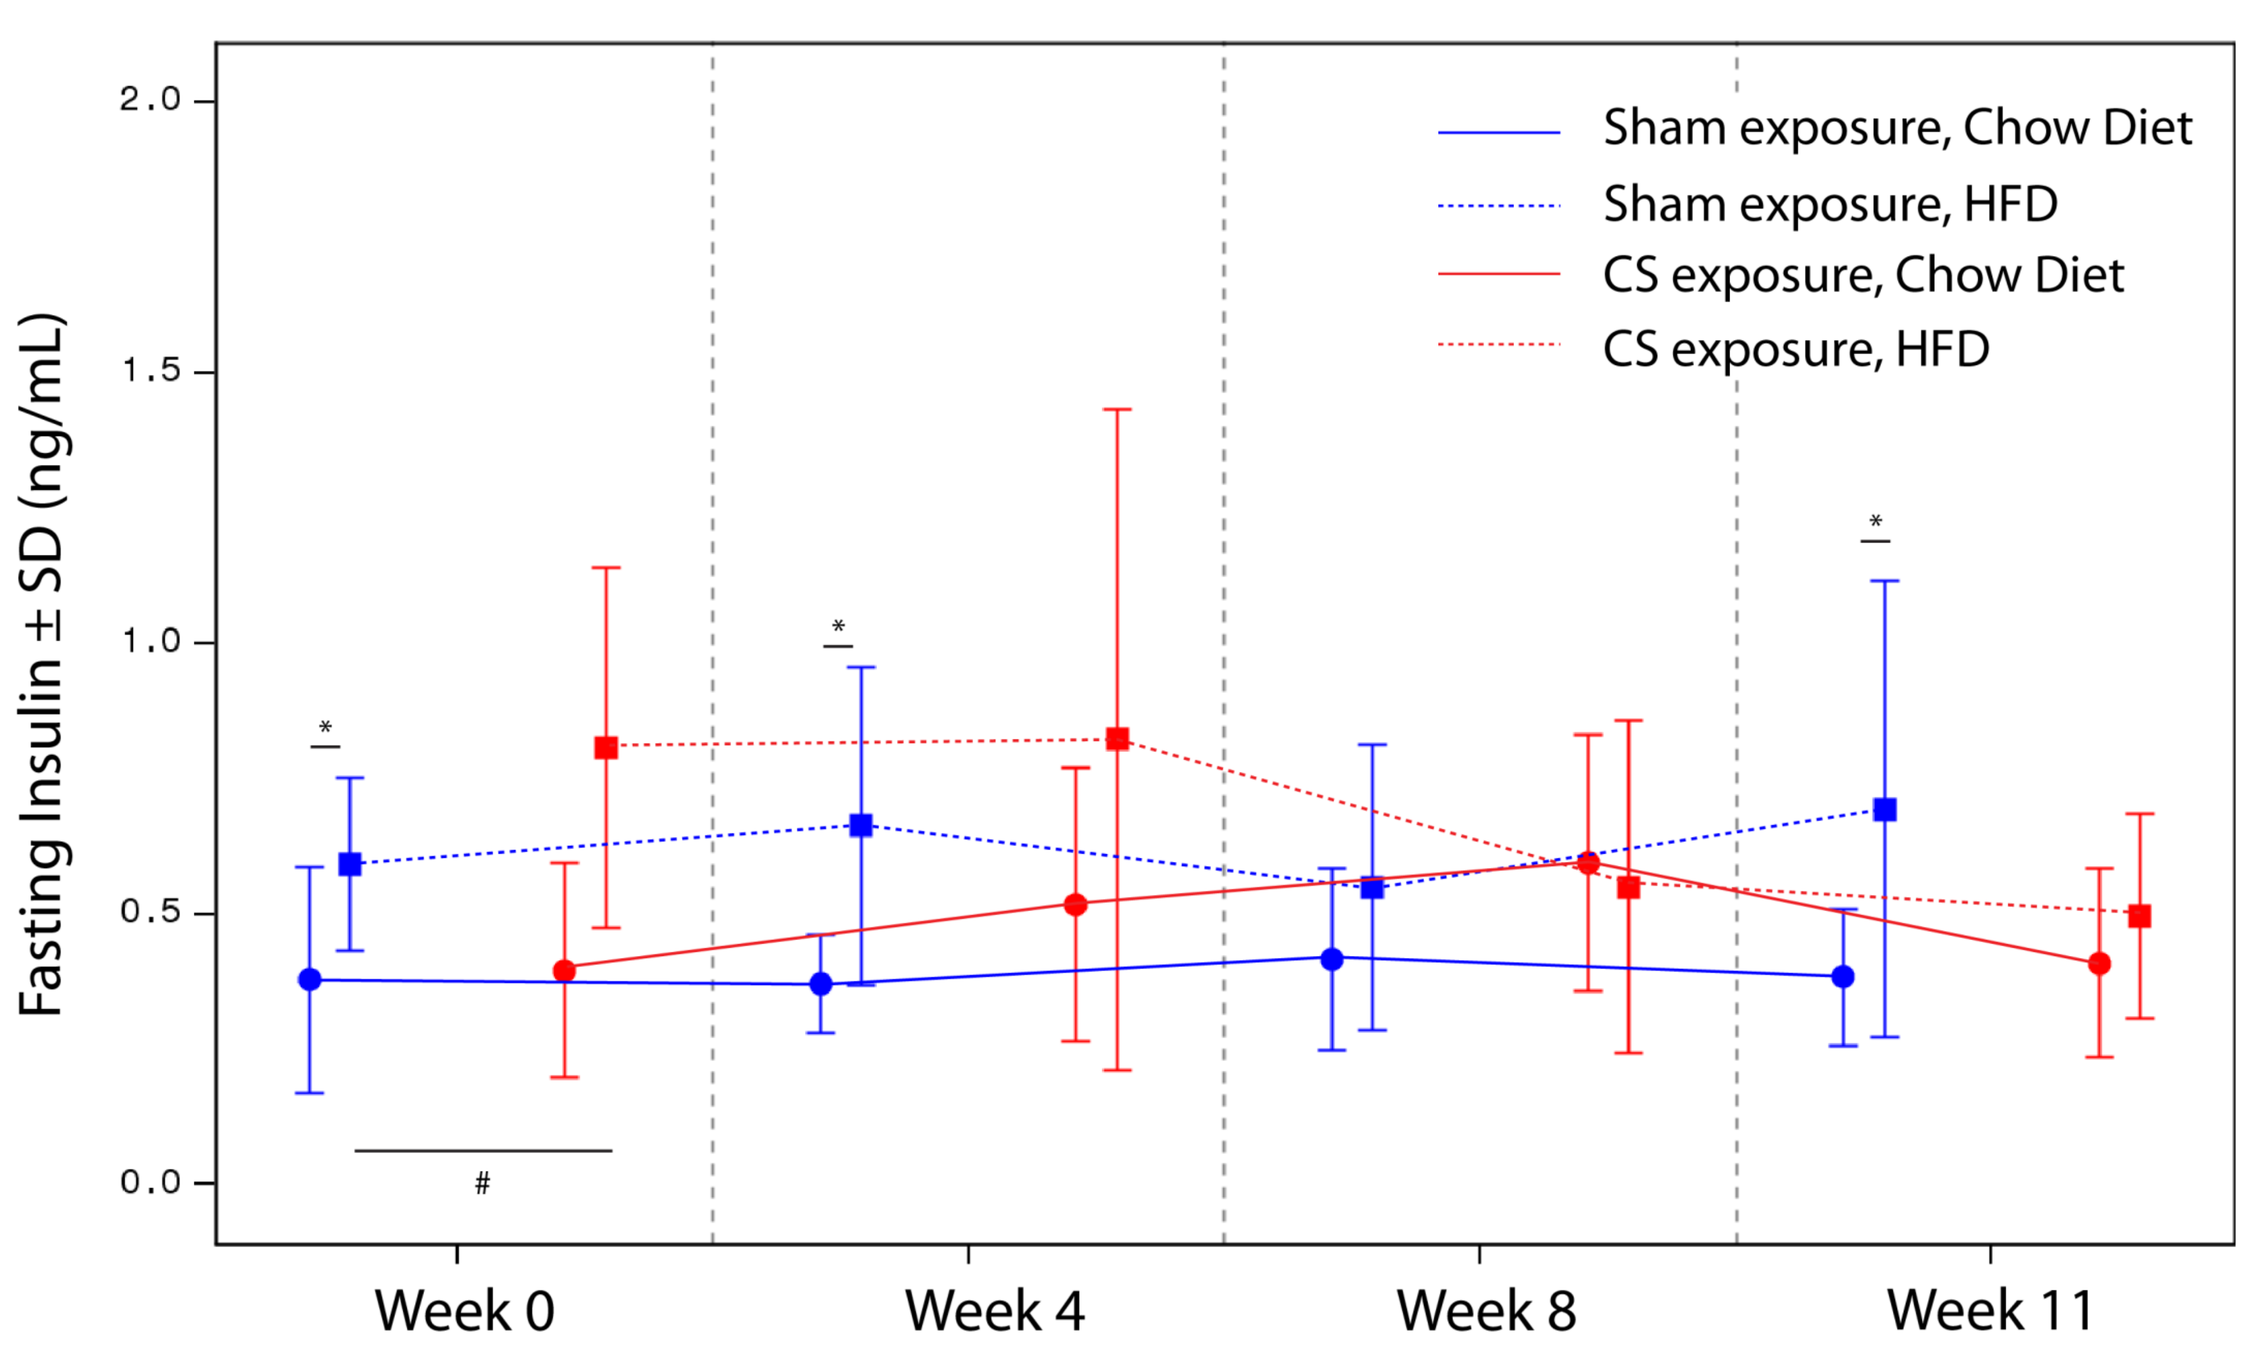
Fasting Insulin ± SD (ng/mL)
Week 0
Week 4
Week 8
Week 11
Sham exposure, Chow Diet
Sham exposure, HFD
CS exposure, Chow Diet
CS exposure, HFD

* $p < 0.05$ as compared to the respective chow-fed group (for the same exposure).
\# $p < 0.05$ between the CS-exposed HFD and sham-exposed HFD.
Measured by a linear model adjusting for the variability between groups.

Blood Lipid Profiles

HFD-fed mice had increased levels of triglycerides, cholesterol, HDL, and LDL as compared with the chow-fed mice. Furthermore, CS exposure significantly increases the lipid levels. Mean ± SD are shown (*$p < 0.05$ ANOVA, Tukey adjustment for multiple comparisons).

Cholesterol

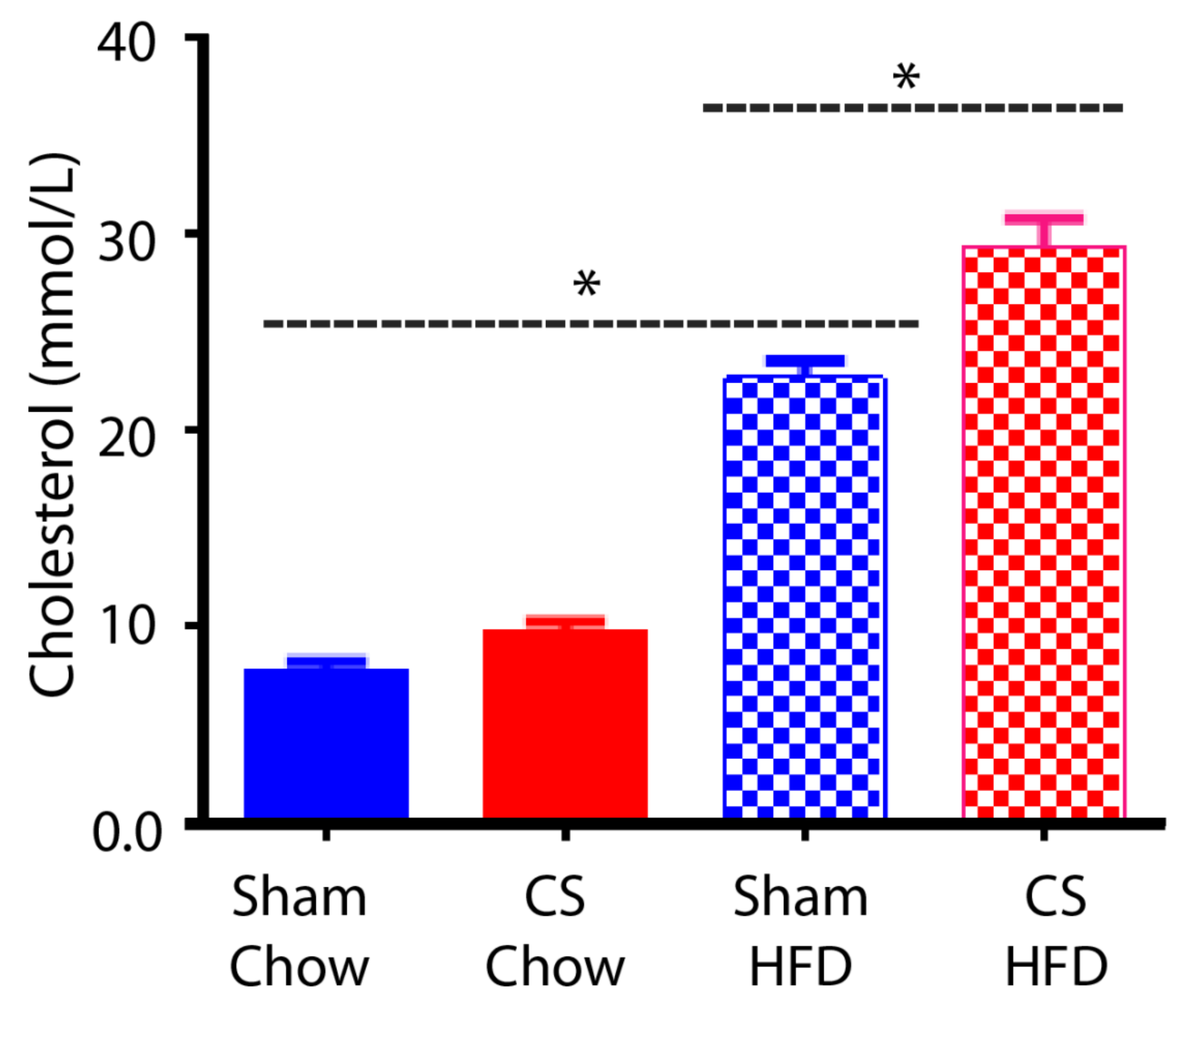
Cholesterol (mmol/L)
Sham Chow
CS Chow
Sham HFD
CS HFD

Triglyceride

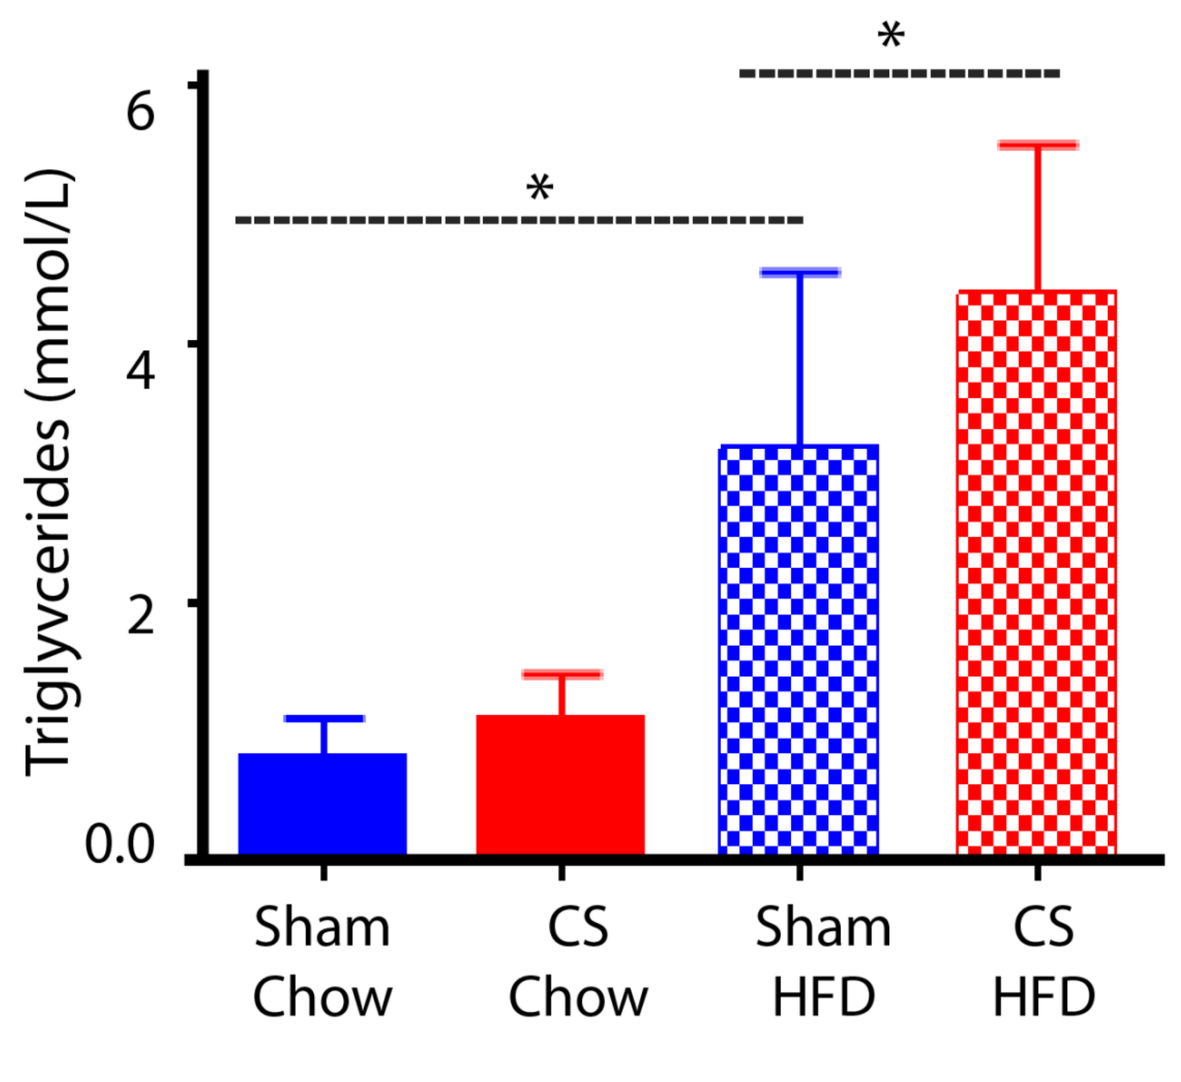
Triglyvcerides (mmol/L)
Sham Chow
CS Chow
Sham HFD
CS HFD

LDL

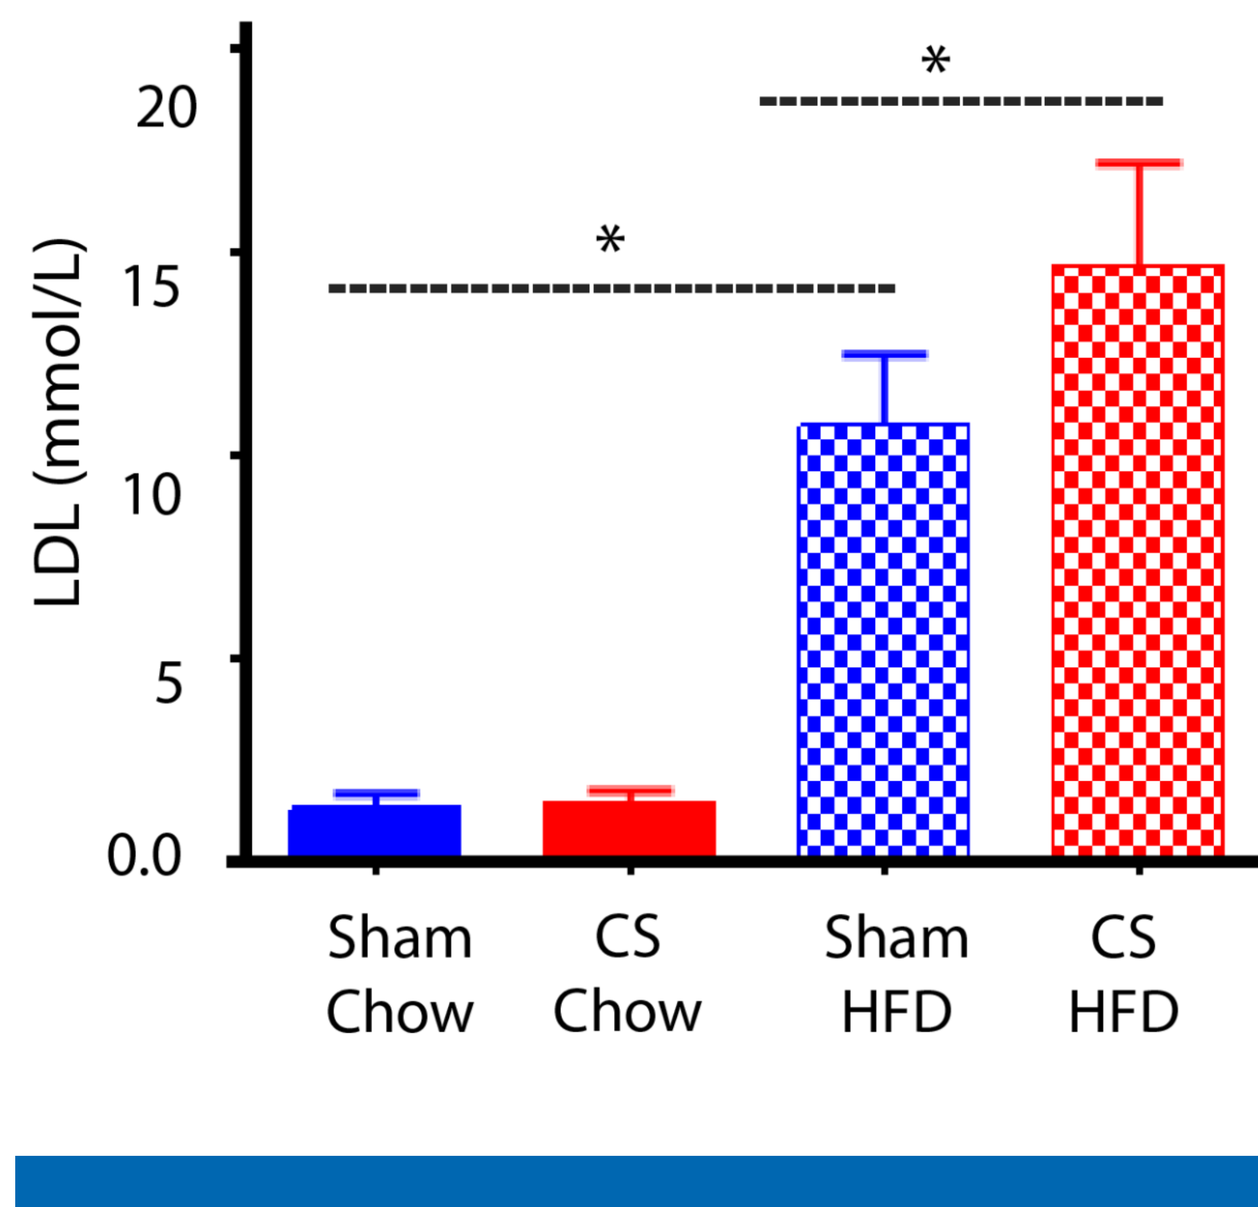
LDL (mmol/L)
Sham Chow
CS Chow
Sham HFD
CS HFD

HDL

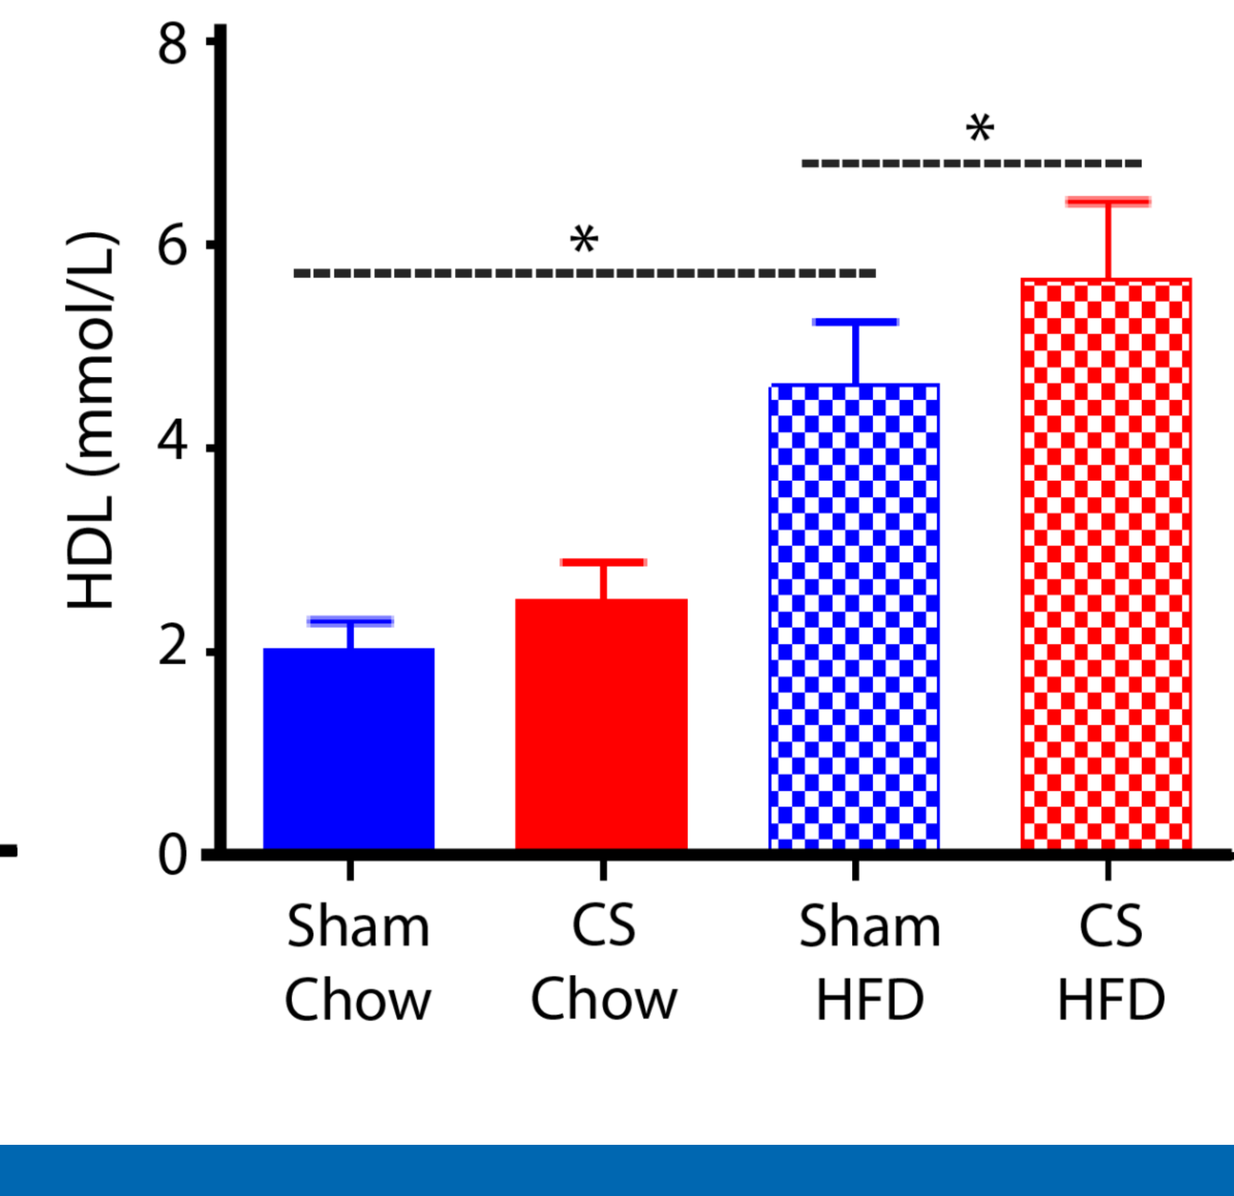
HDL (mmol/L)
Sham Chow
CS Chow
Sham HFD
CS HFD

Serum Metabolic Biomarkers

HFD-fed mice had increased levels of plasminogen activator inhibitor 1 (PAI-1), which has been shown to be correlated with insulin resistance syndromes, as compared with the chow-fed mice. There was a tendency of increased levels of leptin (which has been shown to be elevated in individuals with insulin resistance/type 2 diabetes) and insulin-like growth factor 1 (IGF-1, which has ~50% sequence homology to insulin) in the HFD-fed mice as compared with the chow-fed mice. CS exposure was not associated with alterations of these markers in both chow- and HFD-fed mice. Cortisol levels were increased in the CS-exposed mice only in the chow-fed group, possibly because of the already-increased cortisol that could be induced by HFD . Mean ± SD are shown (*$p < 0.05$ ANOVA, Tukey adjustment for multiple comparisons).

PAI -1

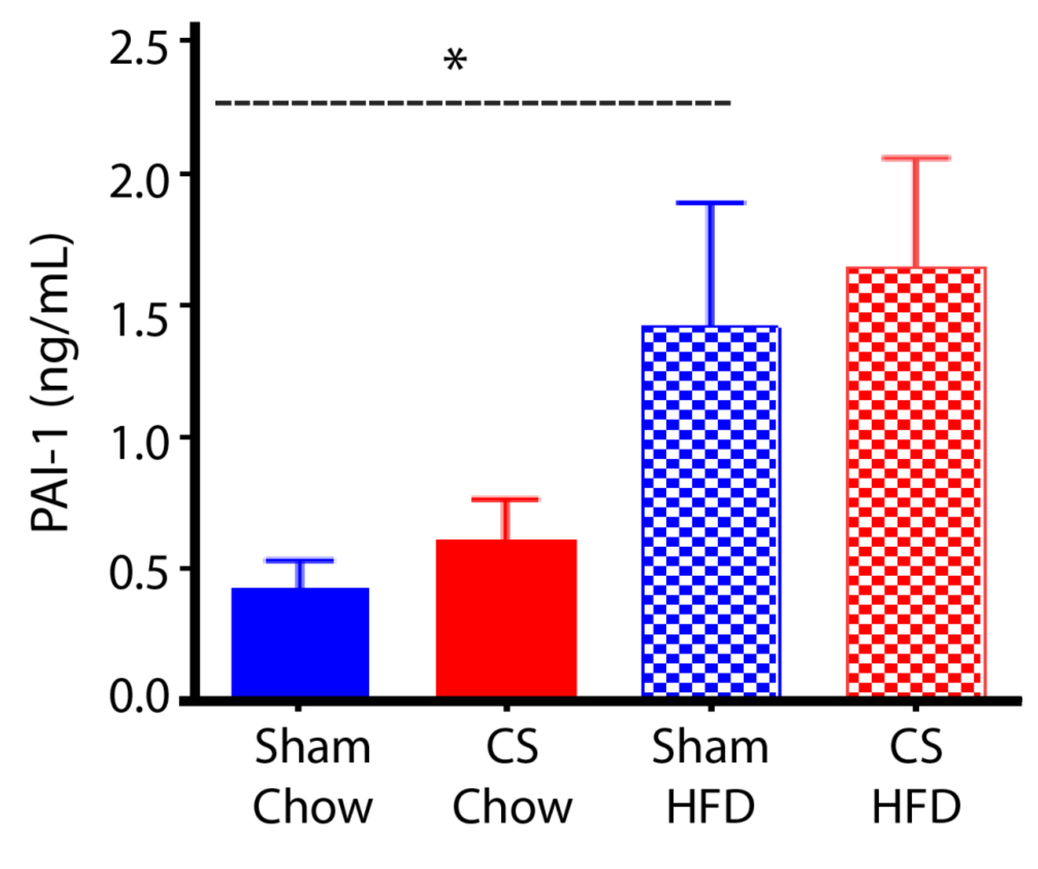
PAI-1 (ng/mL)
Sham Chow
CS Chow
Sham HFD
CS HFD

Leptin

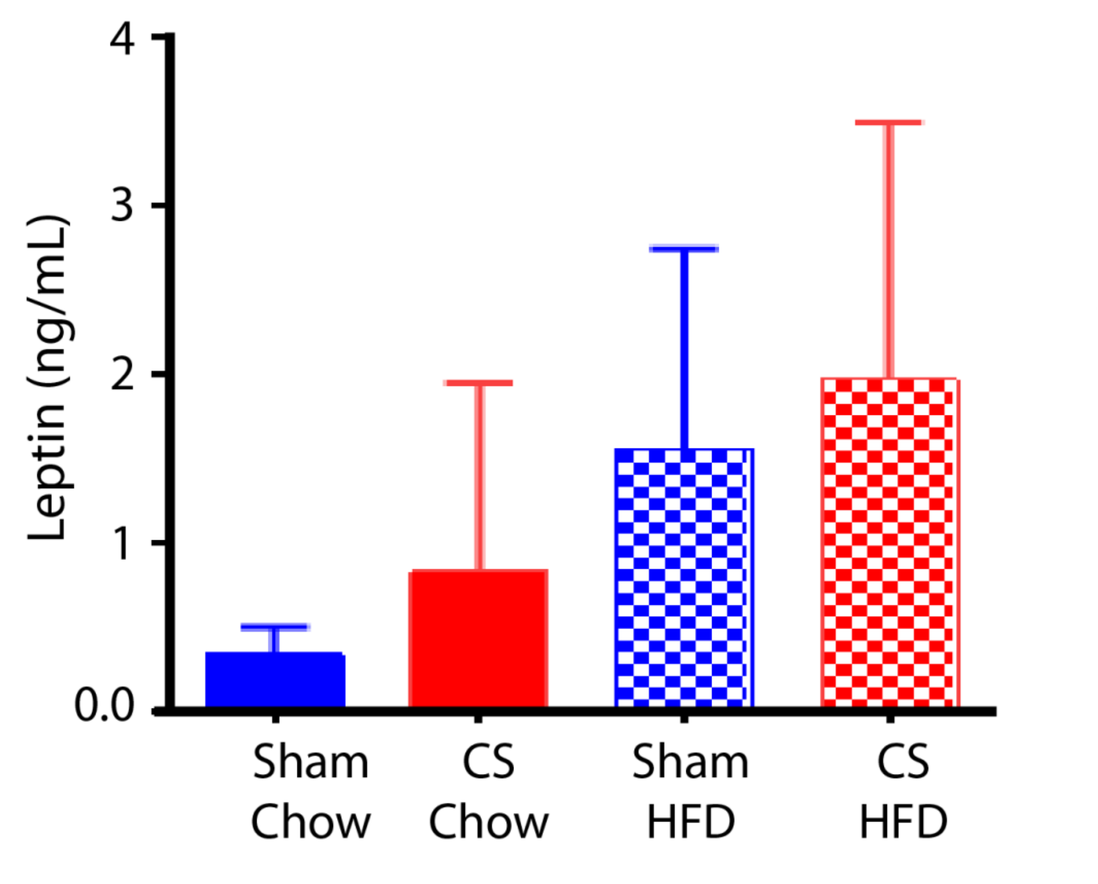
Leptin (ng/mL)
Sham Chow
CS Chow
Sham HFD
CS HFD

IGF-1

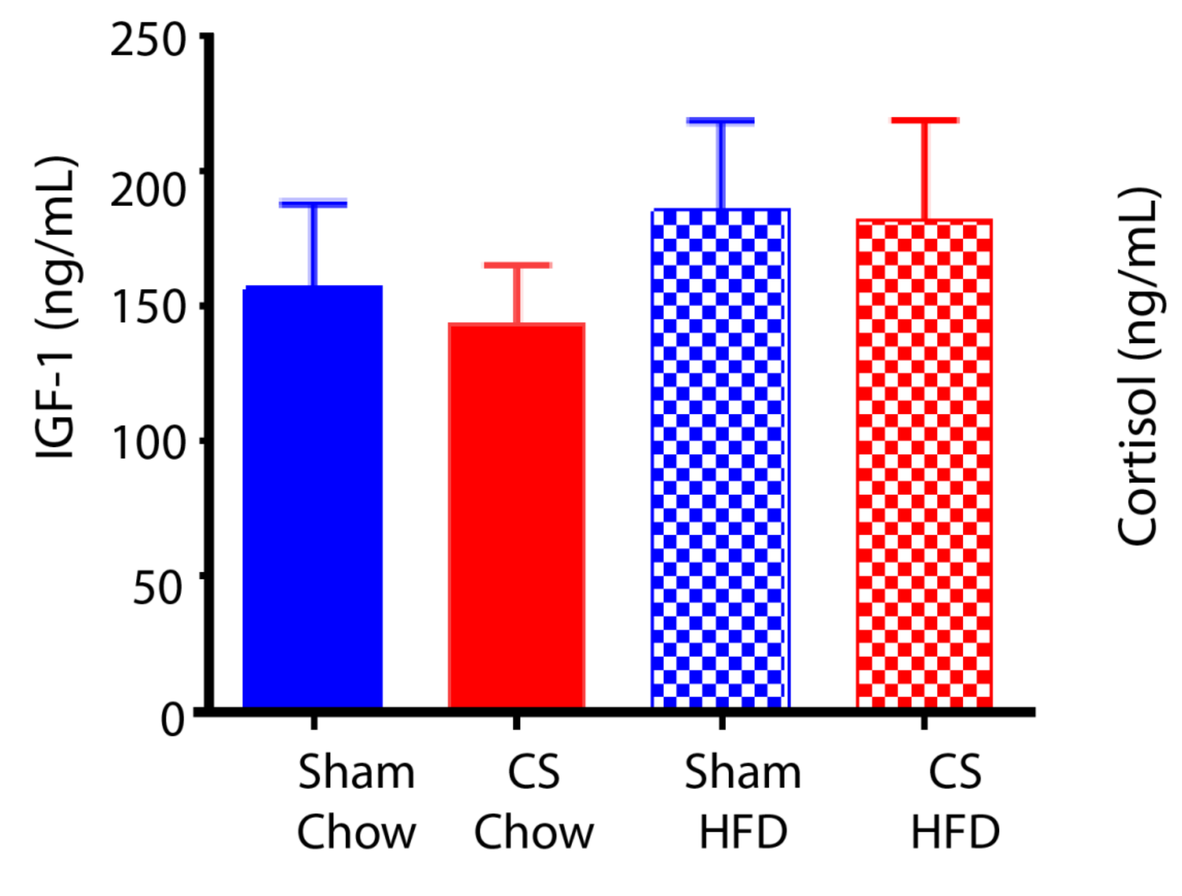
IGF-1 (ng/mL)
Sham Chow
CS Chow
Sham HFD
CS HFD

Cortisol

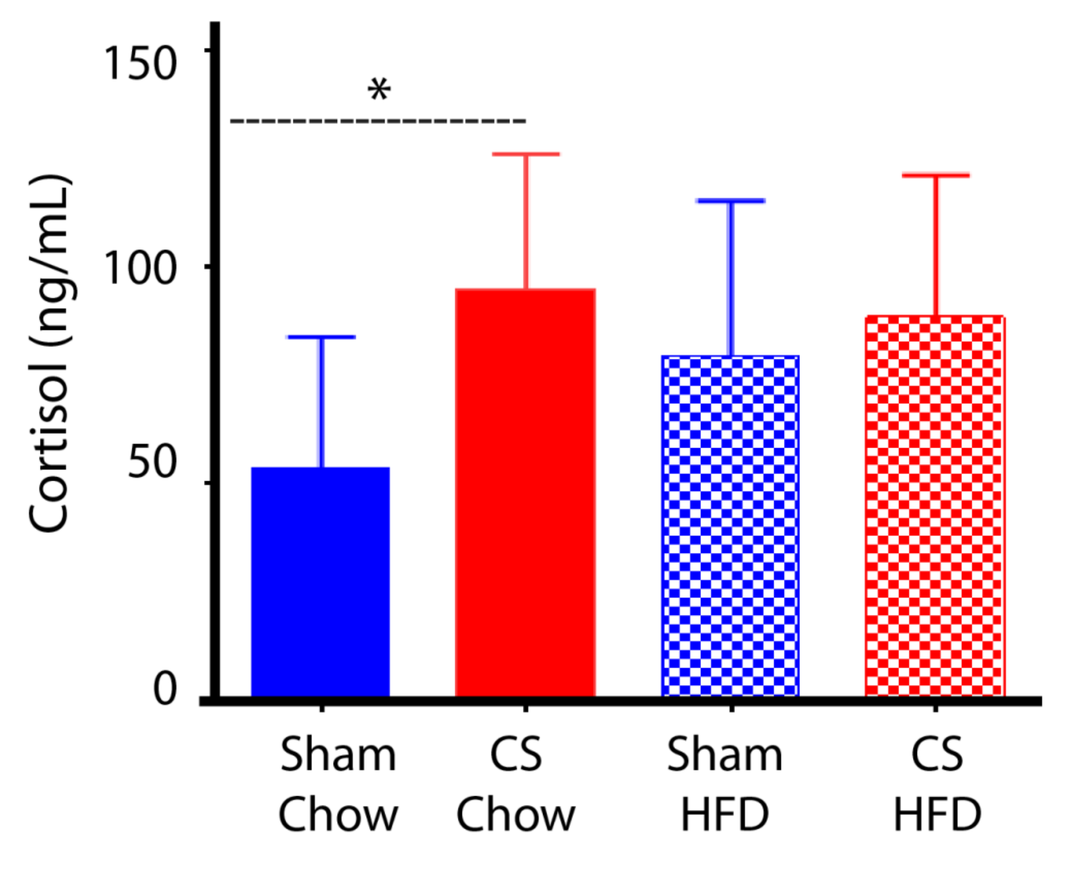
Cortisol (ng/mL)
Sham Chow
CS Chow
Sham HFD
CS HFD

Liver Weight

Greater liver weights were observed in the HFD-fed mice as compared to the chow-fed mice. There was no effect of CS exposure. Mean ± SD are shown (*$p < 0.05$ ANOVA, Tukey adjustment for multiple comparisons).

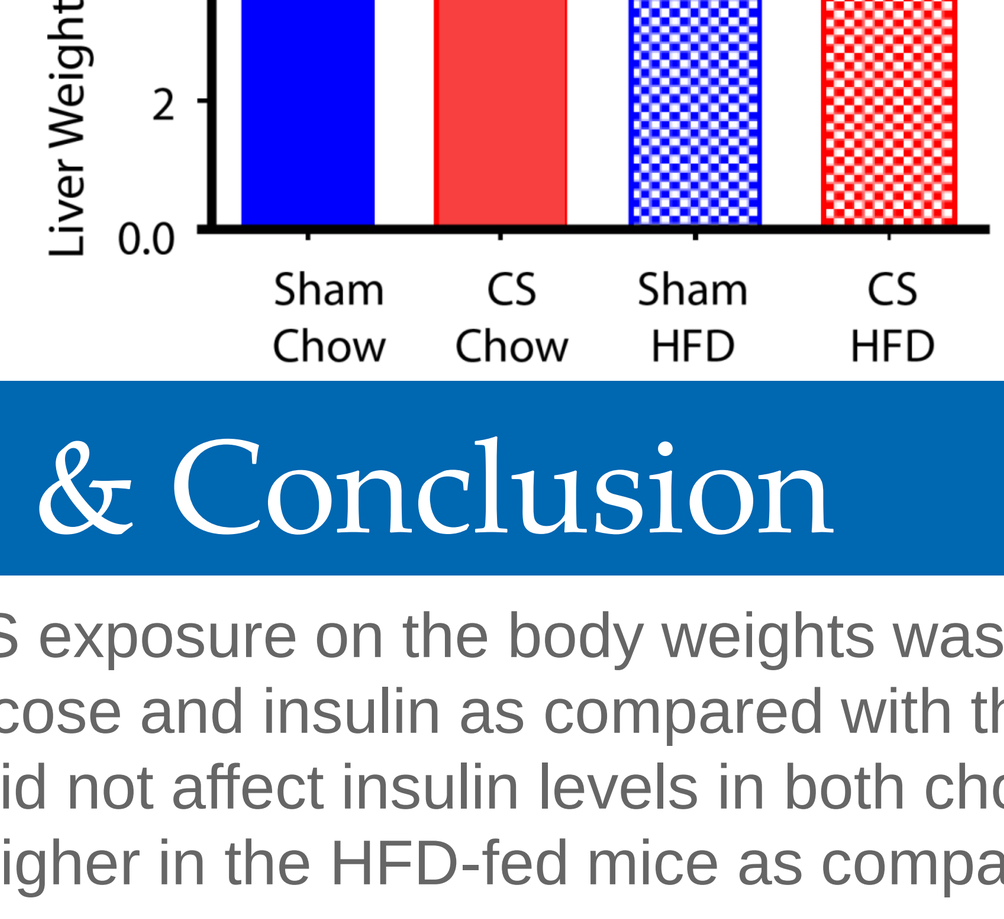
Liver Weight (% BW Normalized)
Sham Chow
CS Chow
Sham HFD
CS HFD

Aortic Plaque Area

Increased plaque area is linked to cardiovascular risk, obesity, and IR. Plaque areas were increased in the HFD-fed mice as compared with the chow-fed mice. Furthermore, exposure of CS significantly increased of plaque area in the HFD-fed mice. Mean ± SD are shown (*$p < 0.05$ ANOVA, Tukey adjustment for multiple comparisons).

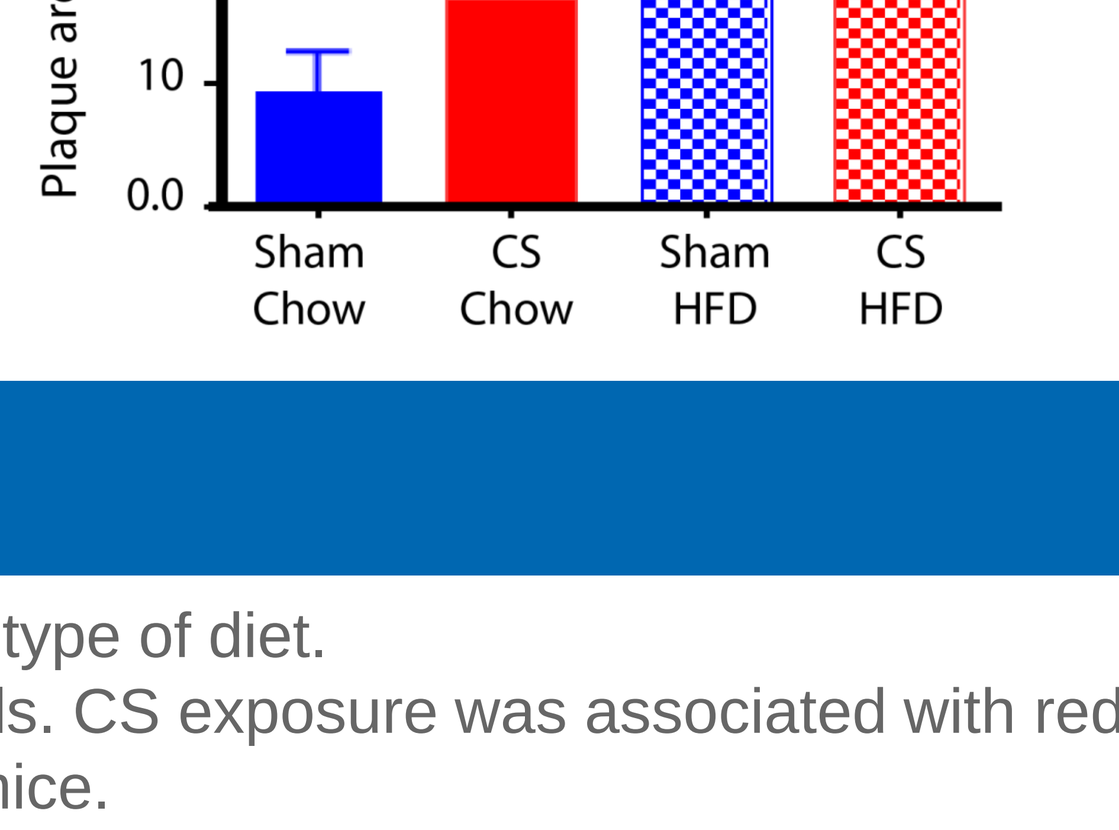
Plaque area (% of Aorta Area)
Sham Chow
CS Chow
Sham HFD
CS HFD

Islet Area

There was a slight increase of islet size in the HFD-fed mice as compared with the chow-fed mice, although this difference was not statistically significant. CS exposure was not associated with alteration of islet are in both chow- and HDS-fed mice. Mean ± SD are shown (*$p < 0.05$ ANOVA, Tukey adjustment for multiple comparisons).

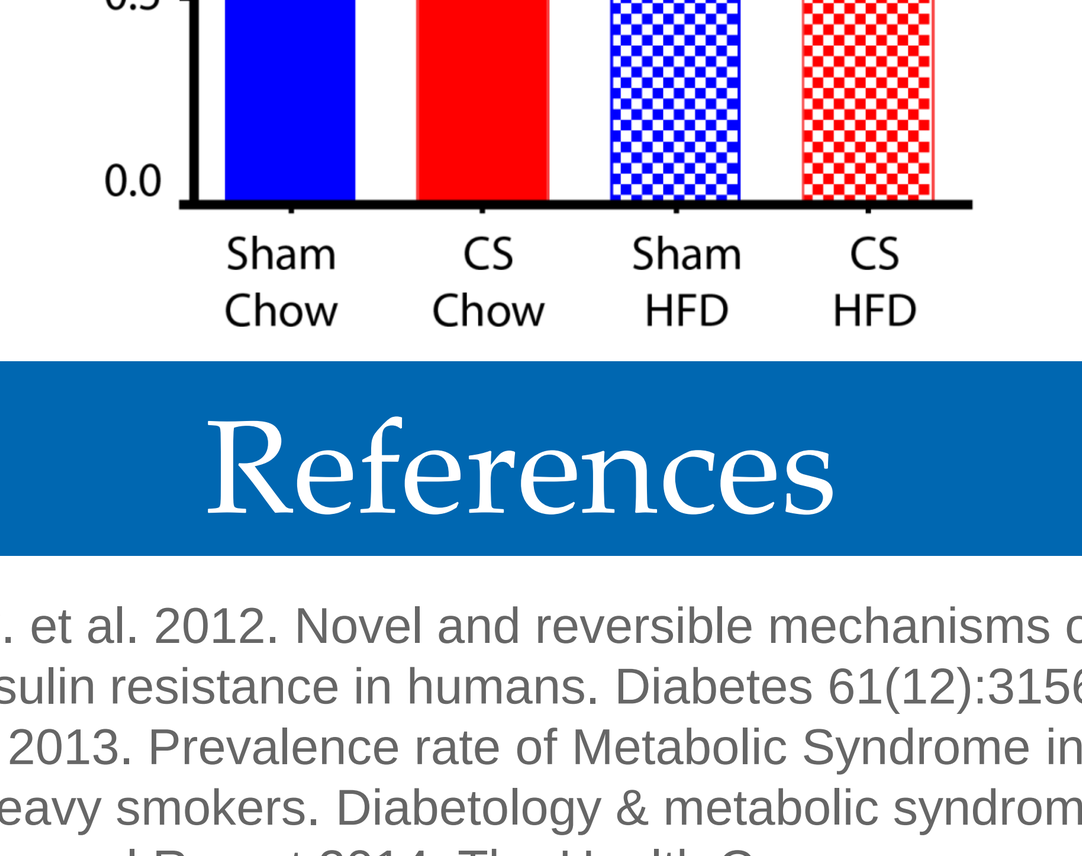
% Islet Area
Sham Chow
CS Chow
Sham HFD
CS HFD

Summary & Conclusion

- HFD-fed mice had greater body weights as compared to the chow-fed mice. The effect of CS exposure on the body weights was dependent on the type of diet.
- At the end of the study (Week 11), the HFD-fed mice had higher levels of fasting plasma glucose and insulin as compared with the chow-fed animals. CS exposure was associated with reduced levels of fasting plasma glucose in both chow- and HFD-fed mice. Moreover, CS exposure did not affect insulin levels in both chow- and HFD-fed mice.
- Levels of cholesterol, triglyceride, HDL, and LDL, as well as plaque area were significantly higher in the HFD-fed mice as compared with the chow-fed mice. Moreover, CS exposure significantly increased the lipid profiles and plaque area in HFD-fed mice.
- HFD-fed mice had greater liver weights and serum levels of PAI-1 as compared with the chow-fed mice; whereas, only trend of increases of serum levels of leptin and IGF-1 were observed. There were no significant effects of CS exposure on the liver weights and serum PAI-1, leptin, and IGF-1 levels in both chow- and HFD-fed mice.
- CS exposure were associated with increased serum cortisol levels only in the chow-fed mice.
- There was a slight increased islet area in the HFD-fed mice as compared to the chow-fed mice, without any effect of the CS exposure.
- In conclusion, although CS exposure was associated with increased blood lipid levels and aortic plaque size in the HFD-fed mice, is was associated with decreased fasting blood glucose levels. This suggests a complex nature of CS effects on glucose and lipid metabolism. The mechanisms by which CS affected glucose and lipid metabolism could be further investigated in the major metabolic organs of the mice using transcriptomics and systems biology approaches, thus allowing further elucidation whether CS exposure in the HFD-fed ApoE-/- mouse model could promote the development of IR.

References

Bergman, B. C. et al. 2012. Novel and reversible mechanisms of smoking-induced insulin resistance in humans. Diabetes 61(12):3156-3166.
Cena, H. et al. 2013. Prevalence rate of Metabolic Syndrome in a group of light and heavy smokers. Diabetology & metabolic syndrome 5:28.
US Surgeon General Report 2014. The Health Consequences of Smoking-50 Years of Progress: A Report of the Surgeon General. US Department of Health & Human Services.
Li, J. et al. 2011. Hyperglycemia in apolipoprotein E-deficient mouse strains with different atherosclerosis susceptibility. Cardiovascular diabetology 10:117.
Noto, H. et al. 2013. Latest insights into the risk of cancer in diabetes. Journal of Diabetes Investigation 4(3):225-232.
Qazi, M. U. and S. Malik. 2013. Diabetes and Cardiovascular Disease: Insights from the Framingham Heart Study. Global Heart 8(1):43-48.
Reference for the 3R4F cigarettes can be found in www.ca.uky.edu/refcig

PMI RESEARCH & DEVELOPMENT

BioPharma ASIA CONVENTION 2014

[1] Philip Morris International R&D, Quai Jeanrenaud 5, CH-2000, Neuchâtel, Switzerland
[2] Phillip Morris International Research Laboratories, Pte Ltd. 50 Science Park Road, Singapore 117400**Philip**