Assessing the impact of switching to the Tobacco Heating System on cardiovascular disease: Translating basic science into clinical benefit

PMI SCIENCE
PHILIP MORRIS INTERNATIONAL

G. Baker, G. de La Bourdonnaye, A. Elamin, C. Goujon, C. Haziza, A. Heremans, J. Hoeng, N. Ivanov, F. Luedicke, S. Maeder, B. Phillips, P. Picavet, S. Pouly, C. Poussin, P. Pratte, C.T. Tran, P. Vanscheeuwijck, M. Peitsch

Presenting/corresponding author: C. Pater

PMI R&D, Morris Products S.A., Quai Jeanrenaud 5, CH-2000, Neuchâtel, Switzerland

Introduction and objectives

Cigarette smoke (CS) is causally linked to the development of cardiovascular diseases (CVD) through different pathophysiologic pathways, including endothelial injury and dysfunction, oxidative stress, procoagulatory status, inflammation, and abnormal lipid profile, all of which contribute to the development of atherosclerosis.

Tobacco harm reduction, by virtue of substituting cigarettes with less harmful products, is a complementary approach to current tobacco control strategies for smokers who would otherwise continue to smoke. The Tobacco Heating System (THS) 2.2 is a novel tobacco product that heats tobacco instead of burning it, never allowing the temperature to exceed 350℃, thereby preventing the combustion process from occurring and producing substantially lower levels of toxicants (on average, >90%) than CS. In particular, the levels of eight cardiovascular toxicants (acrolein, benz(a)anthracene, benzene, butyraldehyde, hydrogen cyanide, lead, phenol, and propionaldehyde) are lower, on average, by >92% in THS aerosol than in CS. Furthermore, THS aerosol does not contain the solid carbon-based nanoparticles (CBNP) that are generated by combustion.

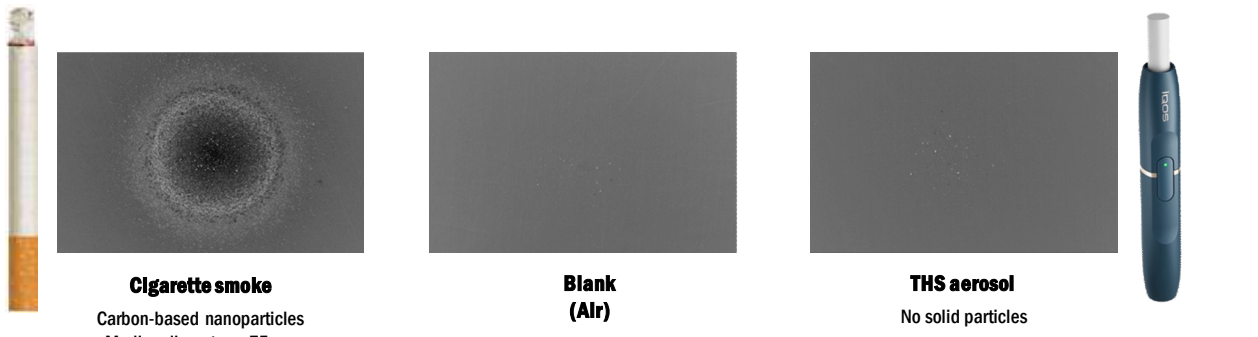
Cigarette smoke
Carbon-based nanoparticles
Median diameter = 75 nm
Amount: 6x10^11 particles =~ 0.7 mg*
Blank
(Air)
THS aerosol
No solid particles

* Under the Health Canada intense smoking regime.

Figure 1. THS generates an aerosol that does not contain CBNPs.

Philip Morris International's (PMI) assessment program aims to demonstrate that switching to THS has the potential to reduce the risk of smoking-related diseases compared with continued smoking. The program includes *in vitro/in vivo* toxicology testing methods that follow Organisation for Economic Co-operation and Development (OECD) and Good Laboratory Practice guidelines and systems toxicology approaches and randomized, controlled clinical studies that follow the principles of Good Clinical Practice.

Methods

Adhesion of monocytes to human coronary arterial endothelial cells (HCAEC), a critical stage in atherosclerosis — THS 2.2 vs. CS (*in vitro* adhesion assay)[1]

Cell exposure to 3R4F reference cigarette or THS 2.2 aqueous smoke/aerosol extract (smoke-/aerosol-bubbled phosphate-buffered saline [PBS; sbPBS/abPBS]).

Conditioned and unconditioned media preparation
Monocytic (MM6) cells were starved in medium for 2 h and then exposed to 3R4F or THS 2.2 sbPBS/abPBS (or PBS) for 2 h. Both types of media were frozen.

Treatment of HCAECs
Indirect and direct treatment: HCAECs were starved for 24 h and then treated with thawed conditioned and unconditioned media for 4 h.
Fresh direct treatment: HCAECs were starved for 24 h and then exposed to freshly generated 3R4F or THS 2.2 sbPBS/abPBS (or PBS) for 4 h. HCAECs and MM6 lysates were collected and stored at –80℃ for RNA extraction.

Adhesion assay
Non-treated MM6 cells and treated (4 h) HCAECs were stained for nuclei for 15 min and then incubated together for 45 min. After cell fixing and washing, the remaining adherent MM6 and HCAEC cells were counted, and the adhesion rate was calculated.

In vivo study to investigate atherosclerotic plaque in the aortic arch[2]

This study examined the development of the hallmarks of CVDs in *Apoe*$^{-/-}$ mice chronically exposed to 3R4F CS, THS 2.2 aerosol (matched to the nicotine concentration in 3R4F CS [30 µg/l]), or filtered air for 3 h per day for 5 days a week for up to 8 months (approximately 40% of their lifetime).

After 2 months of exposure to 3R4F CS, the mice were switched to THS 2.2 aerosol (switching), filtered air (cessation), or continued exposure to 3R4F CS. The exposure dose corresponded to ~30 cigarettes per day in humans.

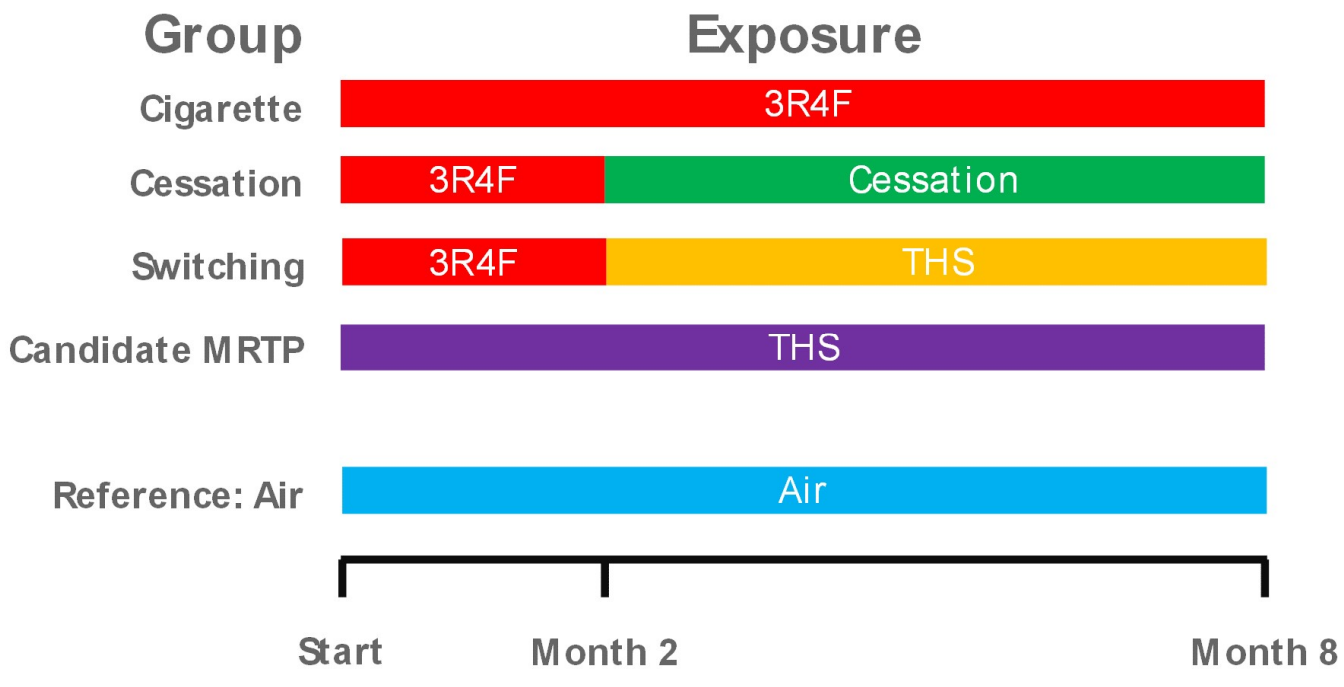
Group
Exposure
Cigarette
3R4F
Cessation
3R4F
Cessation
Switching
3R4F
THS
Candidate MRTP
THS
Reference: Air
Air
Start
Month 2
Month 8

Figure 2: Groups and exposure.

Clinical study — Biological and functional changes in THS switchers[3]

A randomized, controlled, two-arm parallel group, multicenter US study was conducted over 6 months in adult smokers who switched from cigarettes to THS 2.2, in order to demonstrate the favorable changes in THS 2.2 users (≥70%) relative to the changes in those who continued to smoke cigarettes in eight co-primary endpoints representative of the pathomechanistic pathways leading to atherosclerosis (e.g., inflammation, lipid metabolism, endothelial function, platelet function, and oxidative stress). A total of 984 subjects were randomized to the continued cigarette smoking (n = 496) or THS 2.2 (n = 488) groups.

Results

In vitro model — Adhesion assay

- 3R4F sbPBS promoted adhesion of MM6 cells to HCAECs in indirect and fresh direct exposure conditions.
- At the same concentrations, no significant adhesion of MM6 cells to HCAECs was observed following treatment with THS 2.2 abPBS.
- To induce similar effects as 3R4F, THS 2.2 required an approximately 10- to 20-fold increase in concentration.

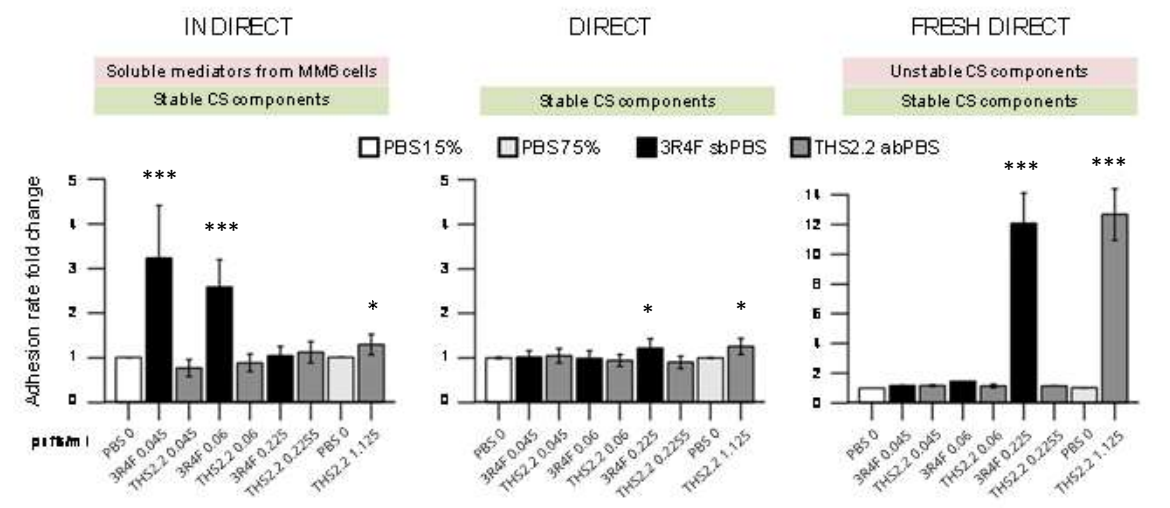
INDIRECT
Soluble mediators from MM6 cells
Stable CS components
DIRECT
Stable CS components
FRESH DIRECT
Unstable CS components
Stable CS components
PBS15%
PBS75%
3R4F sbPBS
THS2.2 abPBS
Adhesion rate fold change
puffs/ml
PBS 0
3R4F 0.045
THS2.2 0.045
3R4F 0.06
THS2.2 0.06
3R4F 0.225
THS2.2 0.2255
PBS 0
THS2.2 1.125

* $p \leq 0.05$, *** $p \leq 0.001$ vs. 0 puffs/ml (PBS 15% or 75%).

Figure 3. Effects of THS 2.2 and 3R4F aqueous extracts on adhesion of MM6 cells to HCAECs following indirect, direct, and fresh direct treatment of HCAECs.

In vivo model: Micro-computed tomography (µCT) data on atherosclerotic plaque in the aortic arch at month 7

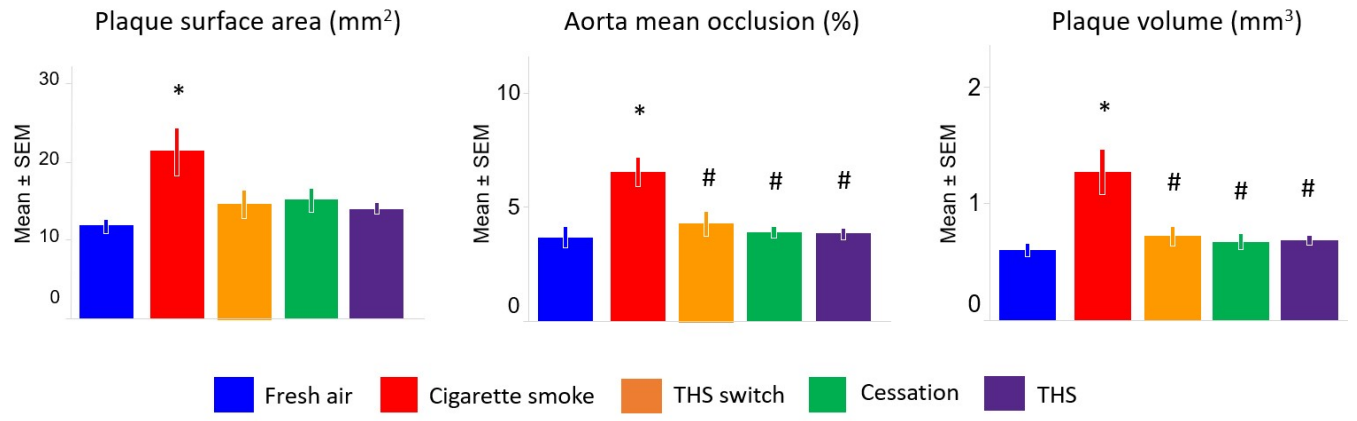
Plaque surface area (mm²)
Aorta mean occlusion (%)
Plaque volume (mm³)
Mean ± SEM
Fresh air
Cigarette smoke
THS switch
Cessation
THS

*Different from sham ($p < 0.05$), #different from CS ($p < 0.05$).

Figure 4. Aortic arch plaque area measurements.

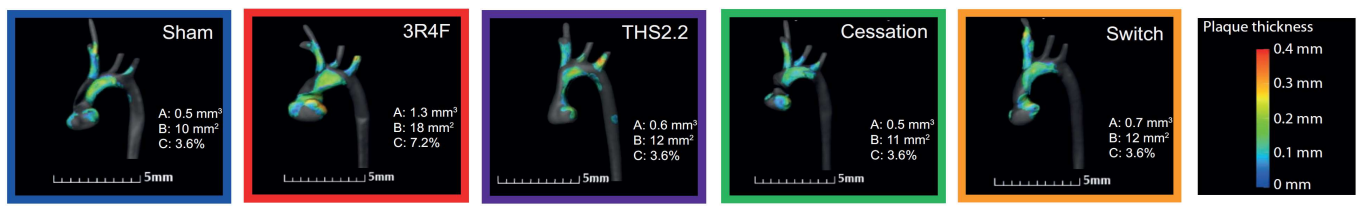
Sham
A: 0.5 mm³
B: 10 mm²
C: 3.6%
3R4F
A: 1.3 mm³
B: 18 mm²
C: 7.2%
THS2.2
A: 0.6 mm³
B: 12 mm²
C: 3.6%
Cessation
A: 0.5 mm³
B: 11 mm²
C: 3.6%
Switch
A: 0.7 mm³
B: 12 mm²
C: 3.6%
5mm
Plaque thickness
0.4 mm
0.3 mm
0.2 mm
0.1 mm
0 mm

Figure 5. µCT images of the aorta after the staining (3D reconstruction showing the position and thickness of the plaque).

A: aorta plaque volume (mm³); B: aorta plaque surface area (mm²); C: aorta mean occlusion (%)

Clinical study — Changes in endpoints at month 6[4]

Endpoint	Change from CC use	Expected change (used for sample size calculation)	Observed change LS mean difference/relative reduction	Hailperin–Rüger - adjusted 96.875% CI	1-sided *p* value (0.0156)	THS 2.2 directional change vs. SA (literature)
HDL-C	Difference	3.3 mg/dL	3.09 mg/dL	1.10, 5.09	<0.001*	✓ Significant
WBC count	Difference	–0.6 GI/L	–0.420 GI/L	–0.717, –0.123	0.001*	✓ Significant
sICAM-1	%Reduction	12%	2.86%	–0.426, 6.04	0.030	✓
11-DTX-B2	%Reduction	18%	4.74%	–7.50, 15.6	0.193	✓
8-epi-$PGF_{2\alpha}$	%Reduction	16%	6.80%	–0.216, 13.3	0.018	✓
COHb	%Reduction	65%	32.2%	24.5, 39.0	<0.001*	✓ Significant

Table 1. Changes in endpoints at month 6.

Conclusions and discussion

The results of the THS 2.2 assessment program demonstrate that:

- THS 2.2 aerosol contains no CBNPs. Additionally, the levels of cardiovascular toxicants are reduced, on average, by >92%.
- Adhesion of monocytic cells to HCAECs *in vitro* is significantly lower following THS 2.2 treatment than after exposure to 3R4F CS.
- Switching to THS 2.2 halted the progression of CS-induced atherosclerotic changes *in vivo*.
- In humans, all coprimary endpoints representative of different pathophysiologic pathways leading to atherosclerosis shifted favorably, in the same direction as the smoking cessation effect reported in the literature, after 6 months of switching from cigarettes to THS 2.2.

PMI has completed 18 non-clinical and 10 clinical studies, including those presented here. The evidence available to date indicates that switching to THS presents less risk of harm and has the potential to reduce the risk of smoking-related diseases, such as CVD.

As a next step, PMI will complement its THS assessment program with cardiovascular outcome studies intended to demonstrate the clinical benefits of switching to THS (e.g., reduction in the risk of cardiovascular death, myocardial infraction, and stroke) over continued smoking.

References

1. C. Poussin et al. "Systems toxicology-based assessment of the candidate modified risk tobacco product THS 2. 2 for the adhesion of monocytic cells to human coronary arterial endothelial cells." Toxicology 339 (2016): 73-86.
2. B. Phillips et al. "An 8-month systems toxicology inhalation/cessation study in Apoe−/− mice to investigate cardiovascular and respiratory exposure effects of a candidate modified risk tobacco product, THS 2.2, compared with conventional cigarettes." Toxicological Sciences 149.2 (2015): 411-432.
3. SM Ansari et al. Rationale and Design for a Randomized, Controlled, Multicenter Study to Evaluate Biological and Functional Changes in Healthy Smokers Switching to the Tobacco Heating System 2.2 Versus Continued Tobacco Smoking, JMIR Res Protoc 2018;7(8):e11294
4. F. Luedicke et al. Effects of switching to a heat-not-burn tobacco product on biologically-relevant biomarkers to assess a candidate modified risk tobacco product: a randomized trial, Cancer Epidemiol Biomarkers Prev July 3 2019

68th CSANZ Annual Scientific Meeting
10-13 December 2020

Competing financial interest: The research described in this poster was sponsored by Philip Morris International.
***Corresponding author: Dr. Calin Pater; E-Mail: calin.pater@pmi.com.**