A lung-on-a-chip platform for improved tissue maturation

David Bovard, Antonin Sandoz, Moran Morelli, Anita Iskandar, Keyur Trivedi, Karsta Luettich, Stefan Frentzel, Julia Hoeng

PMI R&D, Philip Morris Products S.A., Quai Jeanrenaud 5, CH-2000 Neuchâtel, Switzerland

Introduction & experimental design

The preparation of 3D organotypic bronchial or small airway tissues requires the use of primary human bronchial cells, which are first expanded in culture flasks, then seeded on the porous membrane of Transwell® inserts. and finally exposed at the air-liquid interface (ALI) to trigger basal cell differentiation into ciliated and goblet cells **(Figure 1)**.

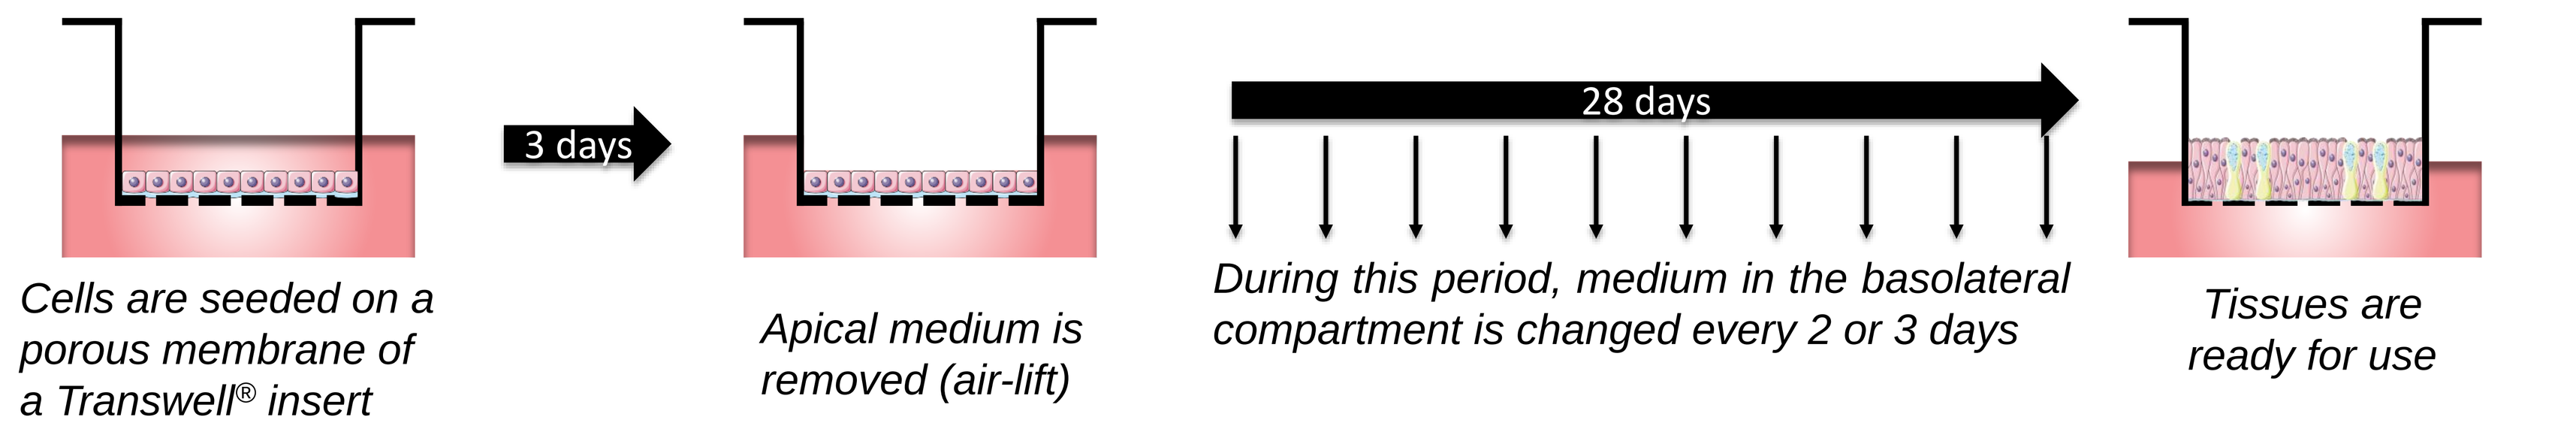
3 days
28 days
Cells are seeded on a porous membrane of a Transwell® insert
Apical medium is removed (air-lift)
During this period, medium in the basolateral compartment is changed every 2 or 3 days
Tissues are ready for use

Figure 1: *Scheme showing the different steps required to mature 3D bronchial tissues at the ALI once the cells have been seeded on the membrane of a Transwell® insert.*

Because the preparation of 3D organotypic bronchial and small airway requires several medium changes over a 4-week period and because the culture in static conditions (where a microenvironment is easily formed, resulting in a non-homogeneous distribution of factors and nutrients) might not be optimal for such tissues, we decided to test whether their maturation could be improved when they are exposed continuously to a medium flow with a lower medium change regimen. Tissues were therefore transferred to a new 32-well chip immediately after the ALI and remained exposed to a continuous medium flow until Day 28 **(Figure 2).** Every week during the maturation, tissues were used to evaluate their characteristics. The results were compared with tissues cultured under standard conditions.

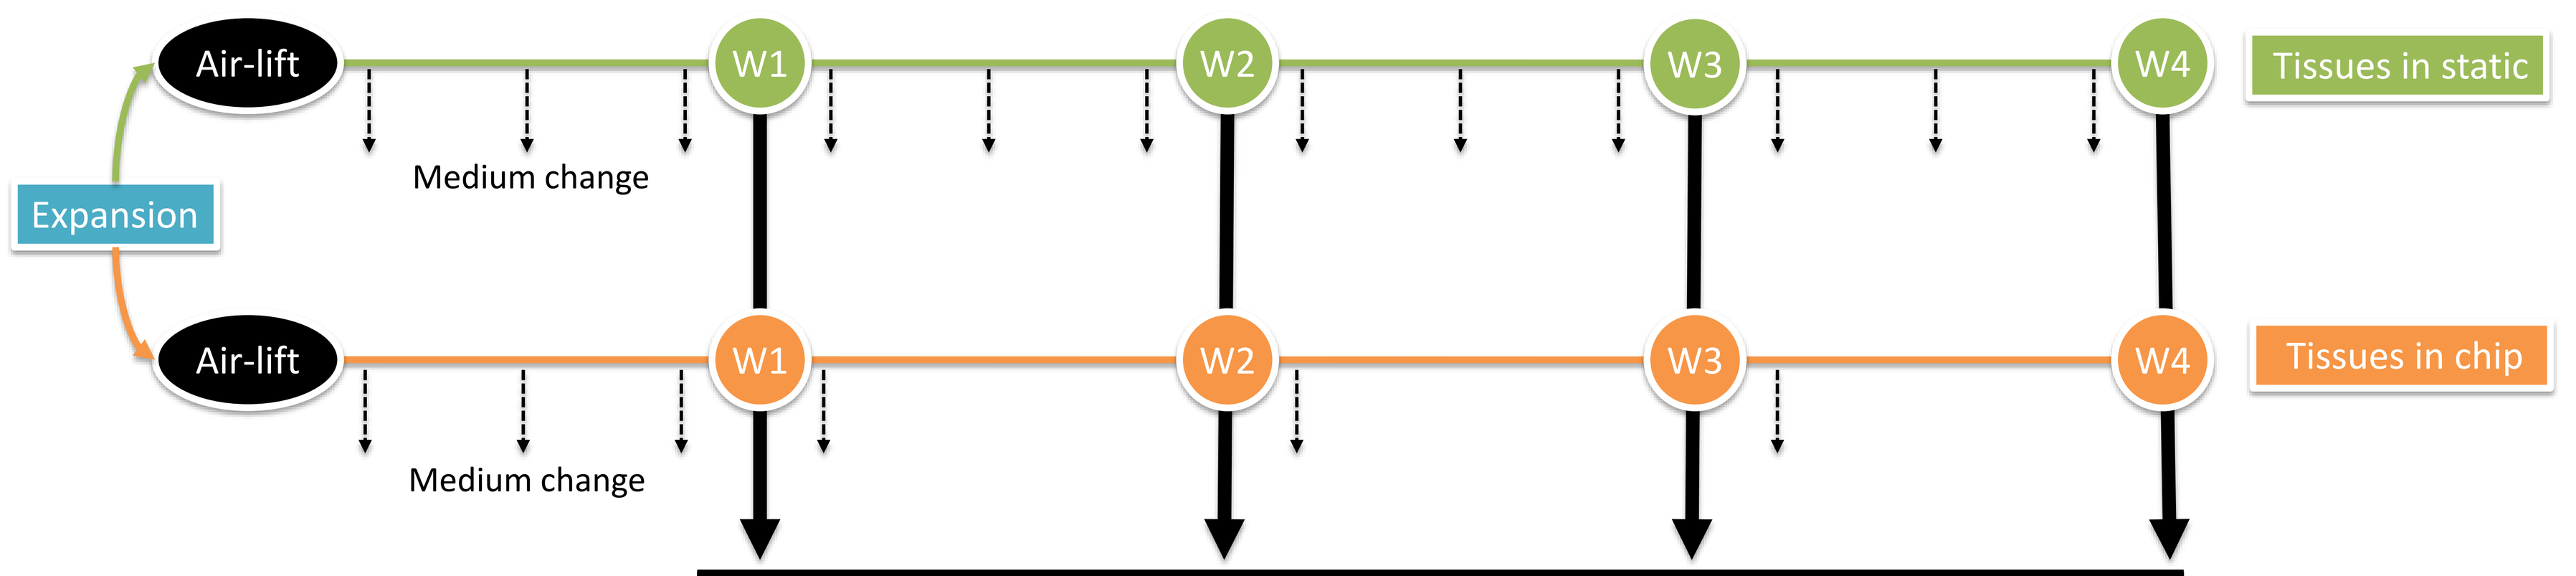
Air-lift
W1
W2
W3
W4
Tissues in static
Medium change
Expansion
Air-lift
W1
W2
W3
W4
Tissues in chip
Medium change

Endpoint	Insert per time-point and tissues type	Longitudinal assay
Histological analysis	3	No
Whole insert staining	4	No
Transepithelial electrical resistance (TEER)	4	Yes
Cilia beating frequency (CBF)	4	Yes
CYP1A1/1B1 enzymatic activity (Week 4 only)	8	No

Figure 2: *Illustration showing the experimental design. Immediately after the air-lift, bronchial tissues (originating from the same cryopreserved vial) were either matured in the static conditions or in the chip. The small dashed arrows show the medium changes. The table below the big arrows shows the assay performed to characterize the tissues every week. CYP = Cytochrome P450; W = Week.*

Methods

Bronchial organotypic cultures were prepared according to the protocol from STEMCELL. Briefly, normal human bronchial epithelial cells (Lonza, Basel, Switzerland) from two different donors were thawed and cultured in a T-flask using PneumaCult™-EX PLUS medium (STEMCELL, Vancouver, Canada) at 37°C with 5% CO_2 and 90% relative humidity. Once the cells were 80% confluent, they were detached from the flask using trypsin-EDTA (Lonza), and 50,000 cells were seeded on a collagen-1-coated Transwell® insert (Ref. 3470, Corning®, Corning, NY, USA). Both apical and basal sides of the cultures were filled with the PneumaCult™-EX PLUS medium and maintained for three days. Subsequently, the culture was air-lifted by removing the apical medium. Half of the cultures were then transferred to the 32-well chip and exposed to a continuous flow of PneumaCult™-ALI medium (8 tissues per circuit, 6 mL of medium per circuit, 150 µL/min), while the other half of the cultures remained in a standard 24-well plate with PneumaCult™-ALI medium in the basolateral compartment.
For the cultures transferred to the 32-well chip, medium was changed every day during the first week and then once a week for the three remaining weeks. For the cultures in static conditions, the medium was changed every 2 or 3 days until Week 4. Tissues were used one week after the transfer for the first week of characterization.
Morphology of bronchial organotypic cultures was evaluated following fixation and paraffin embedding, sectioning, and staining with hematoxylin and eosin (H&E) and Alcian blue. **For the immunostainings**, tissues were first fixed in 4% paraformaldehyde for 20 minutes, then blocked for 1 hour with a solution containing 5% normal goat serum and 1% bovine serum albumin. Then, tissues were stained overnight at 4°C with either an anti β-tubulin 4 antibody conjugated to Alexa 647 (Abcam, Cambridge, UK) or an anti mucin 5AC antibody conjugated to an Alexa 555 (Abcam). Cellular imaging and analysis of the tissues were then acquired using the CellInsight™ CX7 platform (Thermo Fisher Scientific, Waltham, MA, USA)
Transepithelial electrical resistance (TEER) was measured using chopstick electrodes (WPI, Sarasota, FL, USA) connected to an EVOM-2™ Epithelial Voltohmmeter (WPI), according to the manufacturer's instructions.
Cilia beating frequency (CBF) was measured using an inverted microscope (Zeiss, Oberkochen, Germany) equipped with a 4x objective, a chamber warmed at 37°C, and connected to a high-speed camera (Basler AG, Ahrensburg, Germany). Short movies composed of 512 frames recorded at 120 images per second were analyzed using Sisson-Ammons Video Analysis (SAVA) analysis software (Ammons Engineering, Clio, MI, USA).
The metabolic capacity of bronchial tissues was further confirmed by examining the inducibility and activity of CYP1A1/B1 enzymes using P450-Glo™ Assays (Promega, Madison, WI, USA), according to the manufacturer's instructions. CYP1A1/1B1 enzyme activities were induced 48 hours prior to measurement with 10 nM TCDD (Sigma, Buchs, Switzerland).

The technology

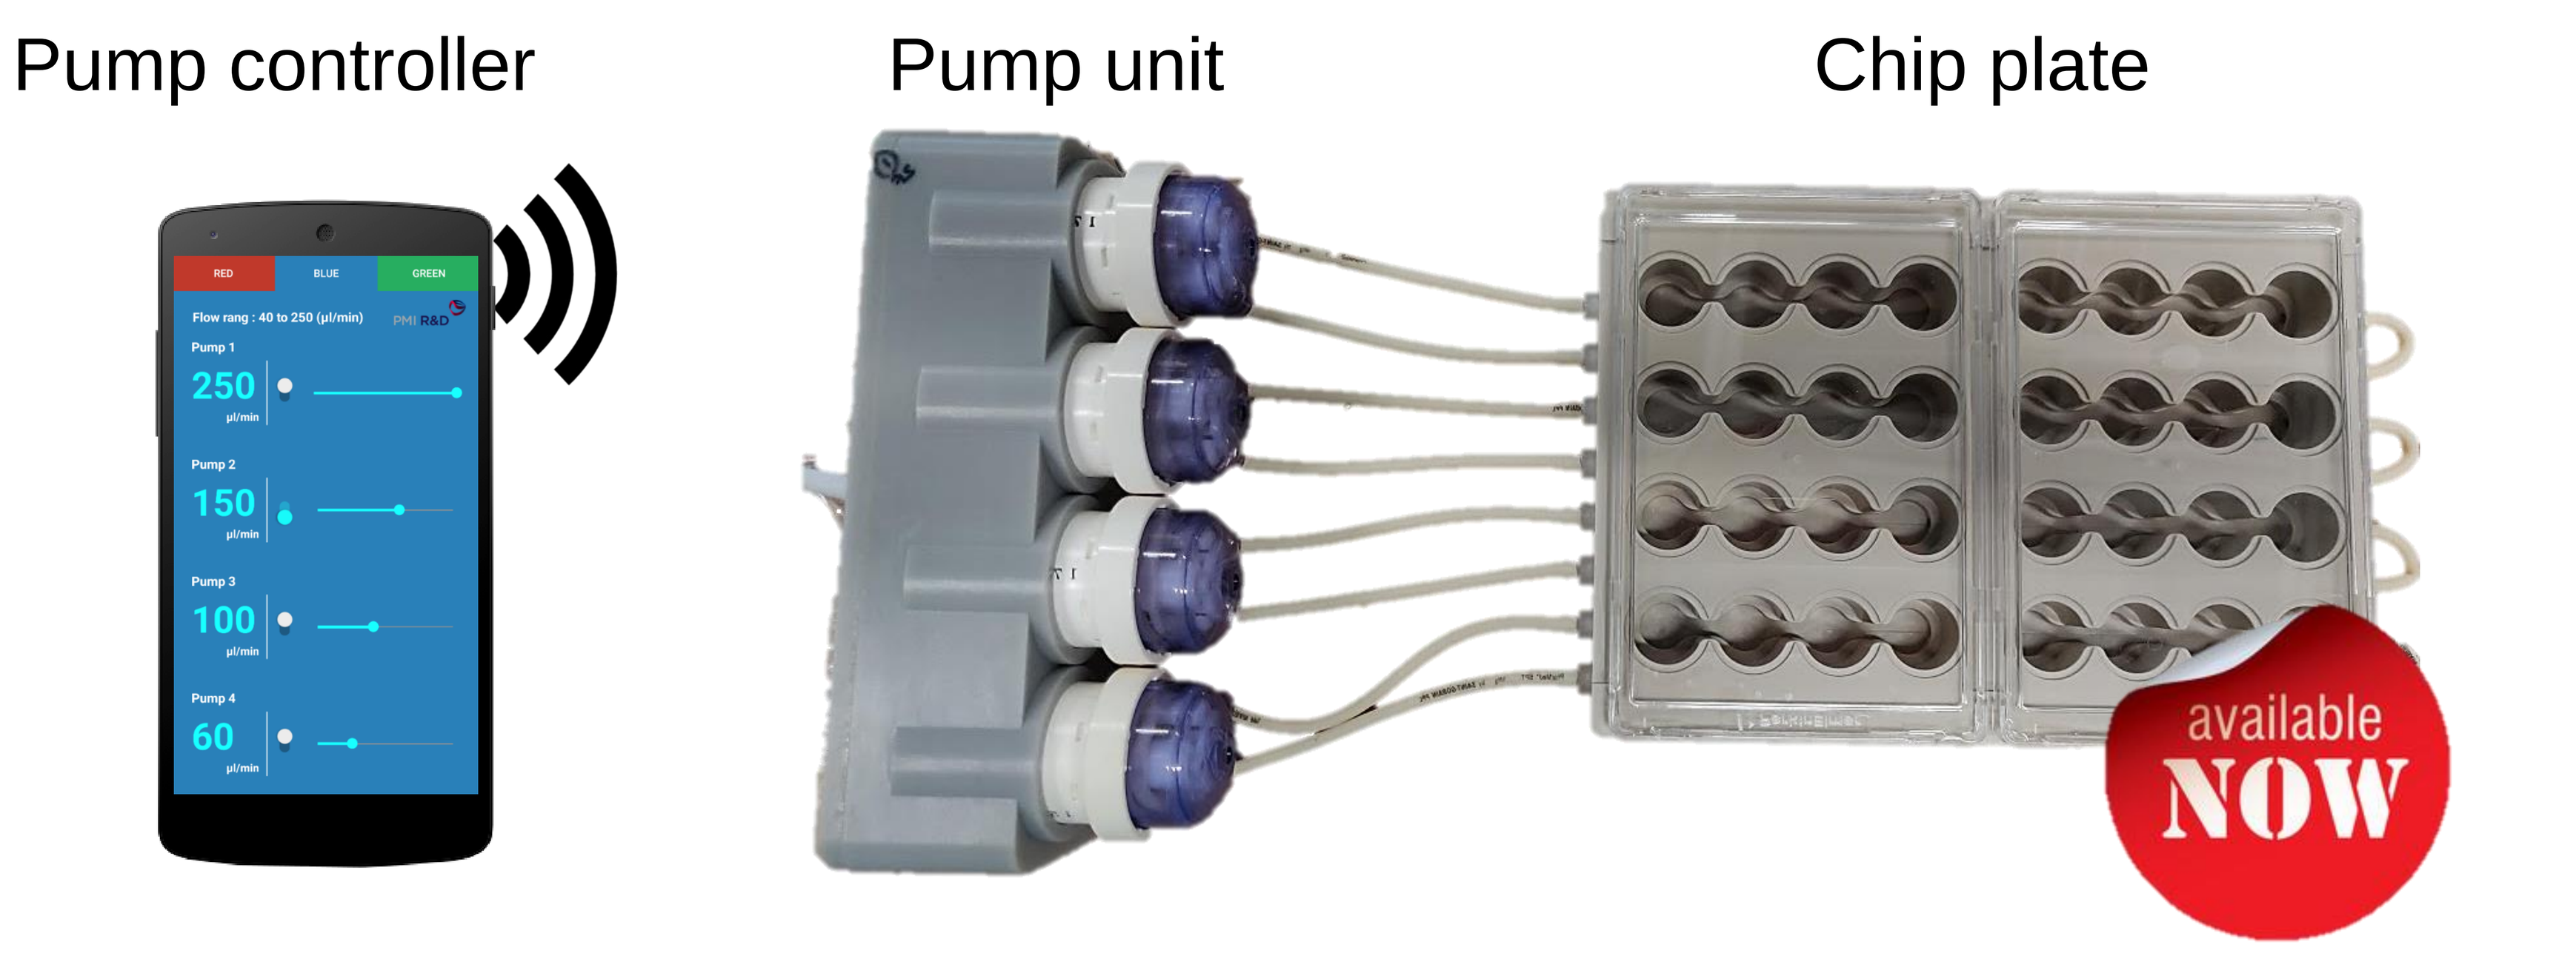
Pump controller
Pump unit
Chip plate
RED
BLUE
GREEN
Flow rang : 40 to 250 (µl/min)
Pump 1
250
Pump 2
150
Pump 3
100
Pump 4
60
available
NOW

Figure 3: *Image showing the chip platform used to improve the maturation of bronchial tissues. The platform is composed of three parts: the smartphone with a specific app used to control the pump rotation speed and direction, the pump unit with four peristaltic pumps, and the 32-well chip plate.*

In order to circulate medium continuously below the Transwell® inserts, we created a new chip plate comprising 32 wells specifically designed for the maturation and maintenance of tissues cultured on Transwell® inserts in a 24-well format.

The new chip plate made from polyetheretherketone has the following features:

- Four circuits, each containing 8 connected wells
- Volume of medium per circuit: 6 to 8 mL

Each circuit of the chip plate is connected individually to the pump unit. The medium flow speed and rotation direction can be individually set using the pump controller. The complete chip platform offers the following essential features:

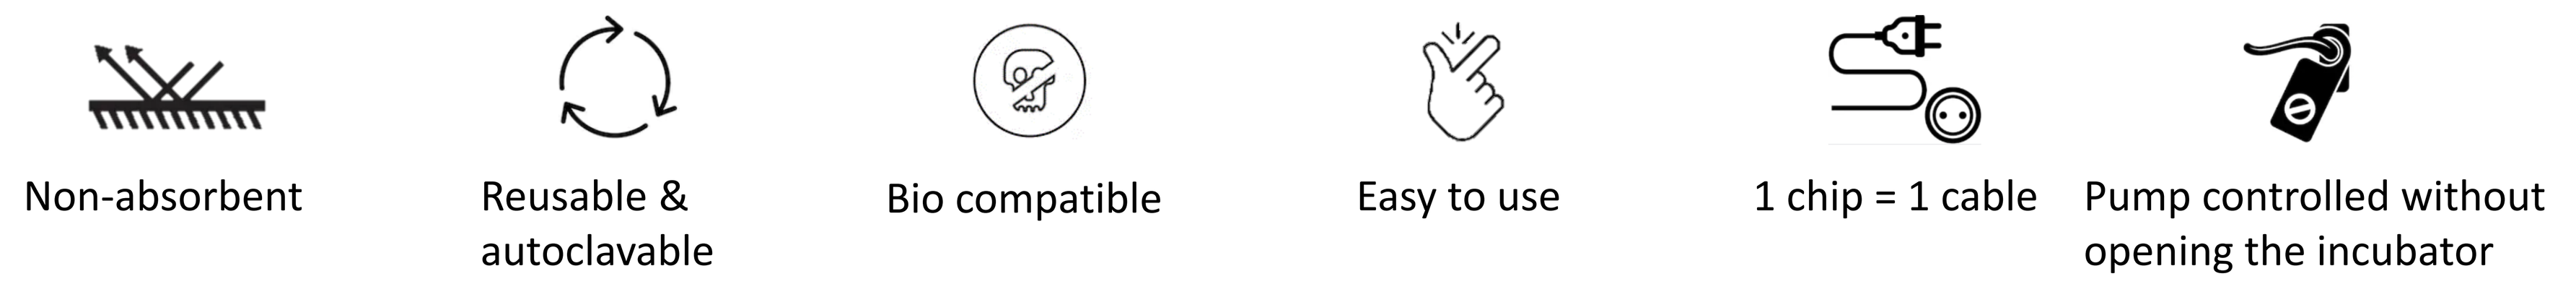
Non-absorbent
Reusable & autoclavable
Bio compatible
Easy to use
1 chip = 1 cable
Pump controlled without opening the incubator

Results

The impact of tissue maturation within the chip was first evaluated by measuring functional parameters of the tissues. The CBF was observed to be slightly lower for tissues maintained in the chip compared to tissues matured in standard conditions **(Figure 4A).** A strong difference in TEER was observed only for the donor 2 at Week 2 between the two conditions; other values measured were similar **(Figure 4B)**. CYP1A1/1B1 activity in tissues, which was only measured at Week 4, was similar for both conditions and both donors. A ~30% decrease in CYP1A1/1B1 induced activity was measured for tissues matured in the chip compared to tissues matured in standard conditions **(Figure 4C)**.
Interestingly, a comparison of tissue morphology between conditions showed striking differences: starting from Week 2 for both donors, the ciliated cells showed a more polarized morphology for tissues maintained in the chip than in tissues matured in static conditions **(Figure 5A)**. At Week 2 and Week 3, tissues matured in the chip were also between 10% and 50%, although this was donor-dependent **(Figure 5B)**. A staining for cilia and goblet cells showed that the tissues matured in the chip had a denser ciliation **(Figure 6)** but a 10% to 50% lower number of goblet cells **(Figure 7)**.

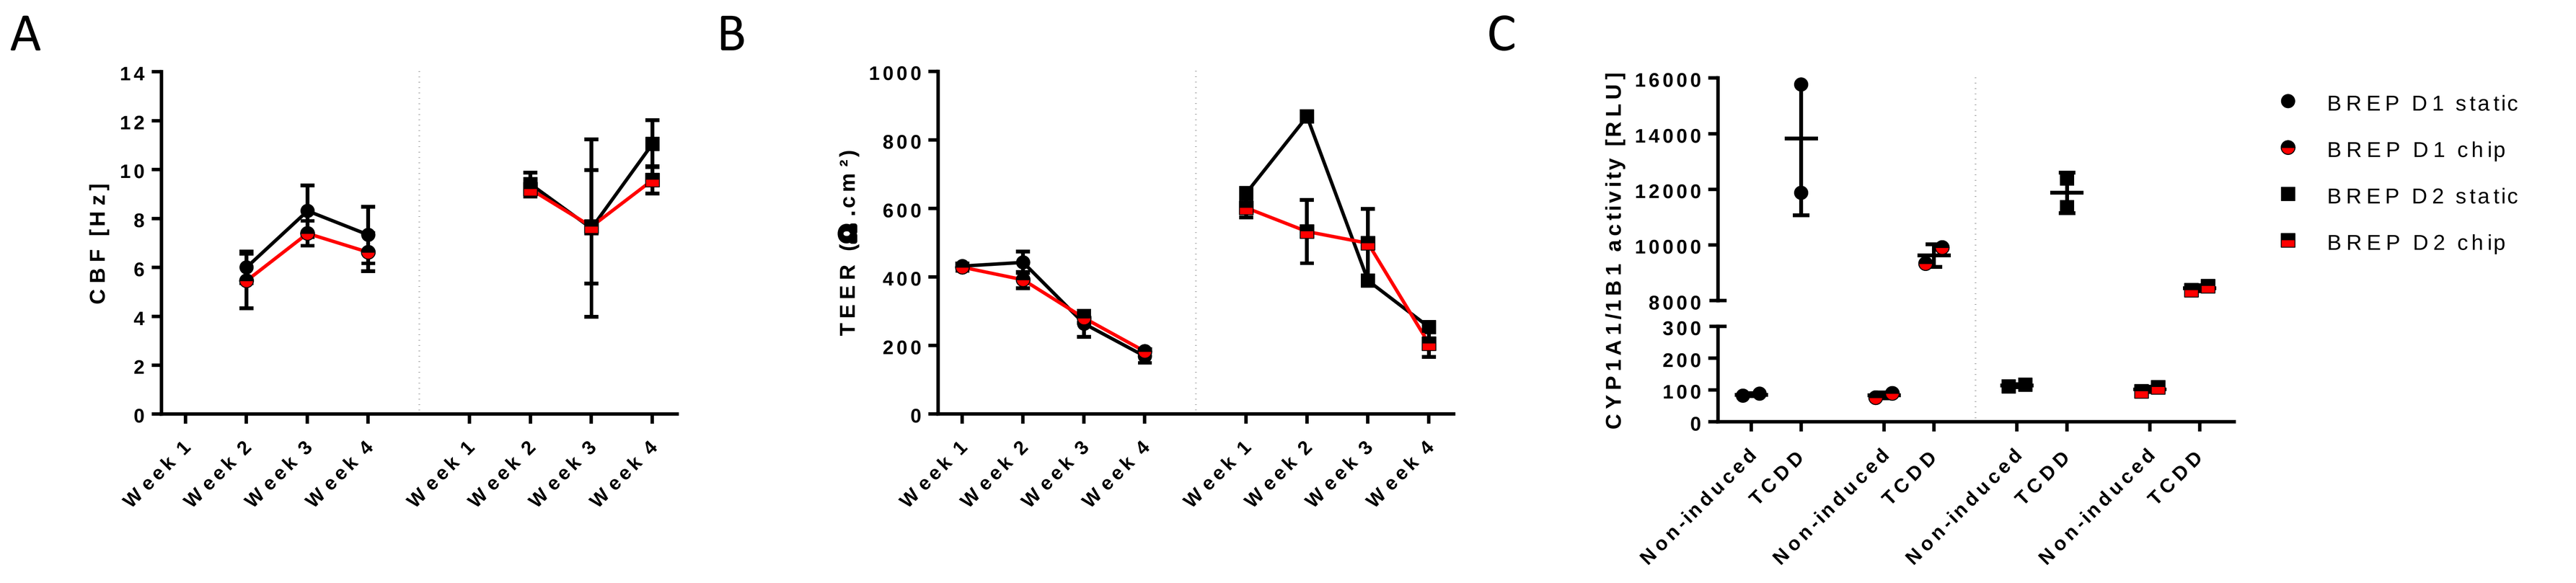
A
B
C
CBF [Hz]
TEER (Ω.cm²)
CYP1A1/1B1 activity [RLU]
Week 1
Week 2
Week 3
Week 4
Non-induced
TCDD
BREP D1 static
BREP D1 chip
BREP D2 static
BREP D2 chip

Figure 4: ***A)*** *CBF and,* ***B)*** *TEER measurement were performed every week on the same 4 tissues.* ***C)*** *CYP1A1/1B1 activity was measured in 2 untreated tissues (non-induced) and in 2 tissues treated with 10 nM TCDD on Week 4. The graphs show the mean ± standard deviation. Hz = Herz; TCDD = 2,3,7,8-Tetrachlordibenzodioxin; RLU = Relative luminescence unit; D = donor.*

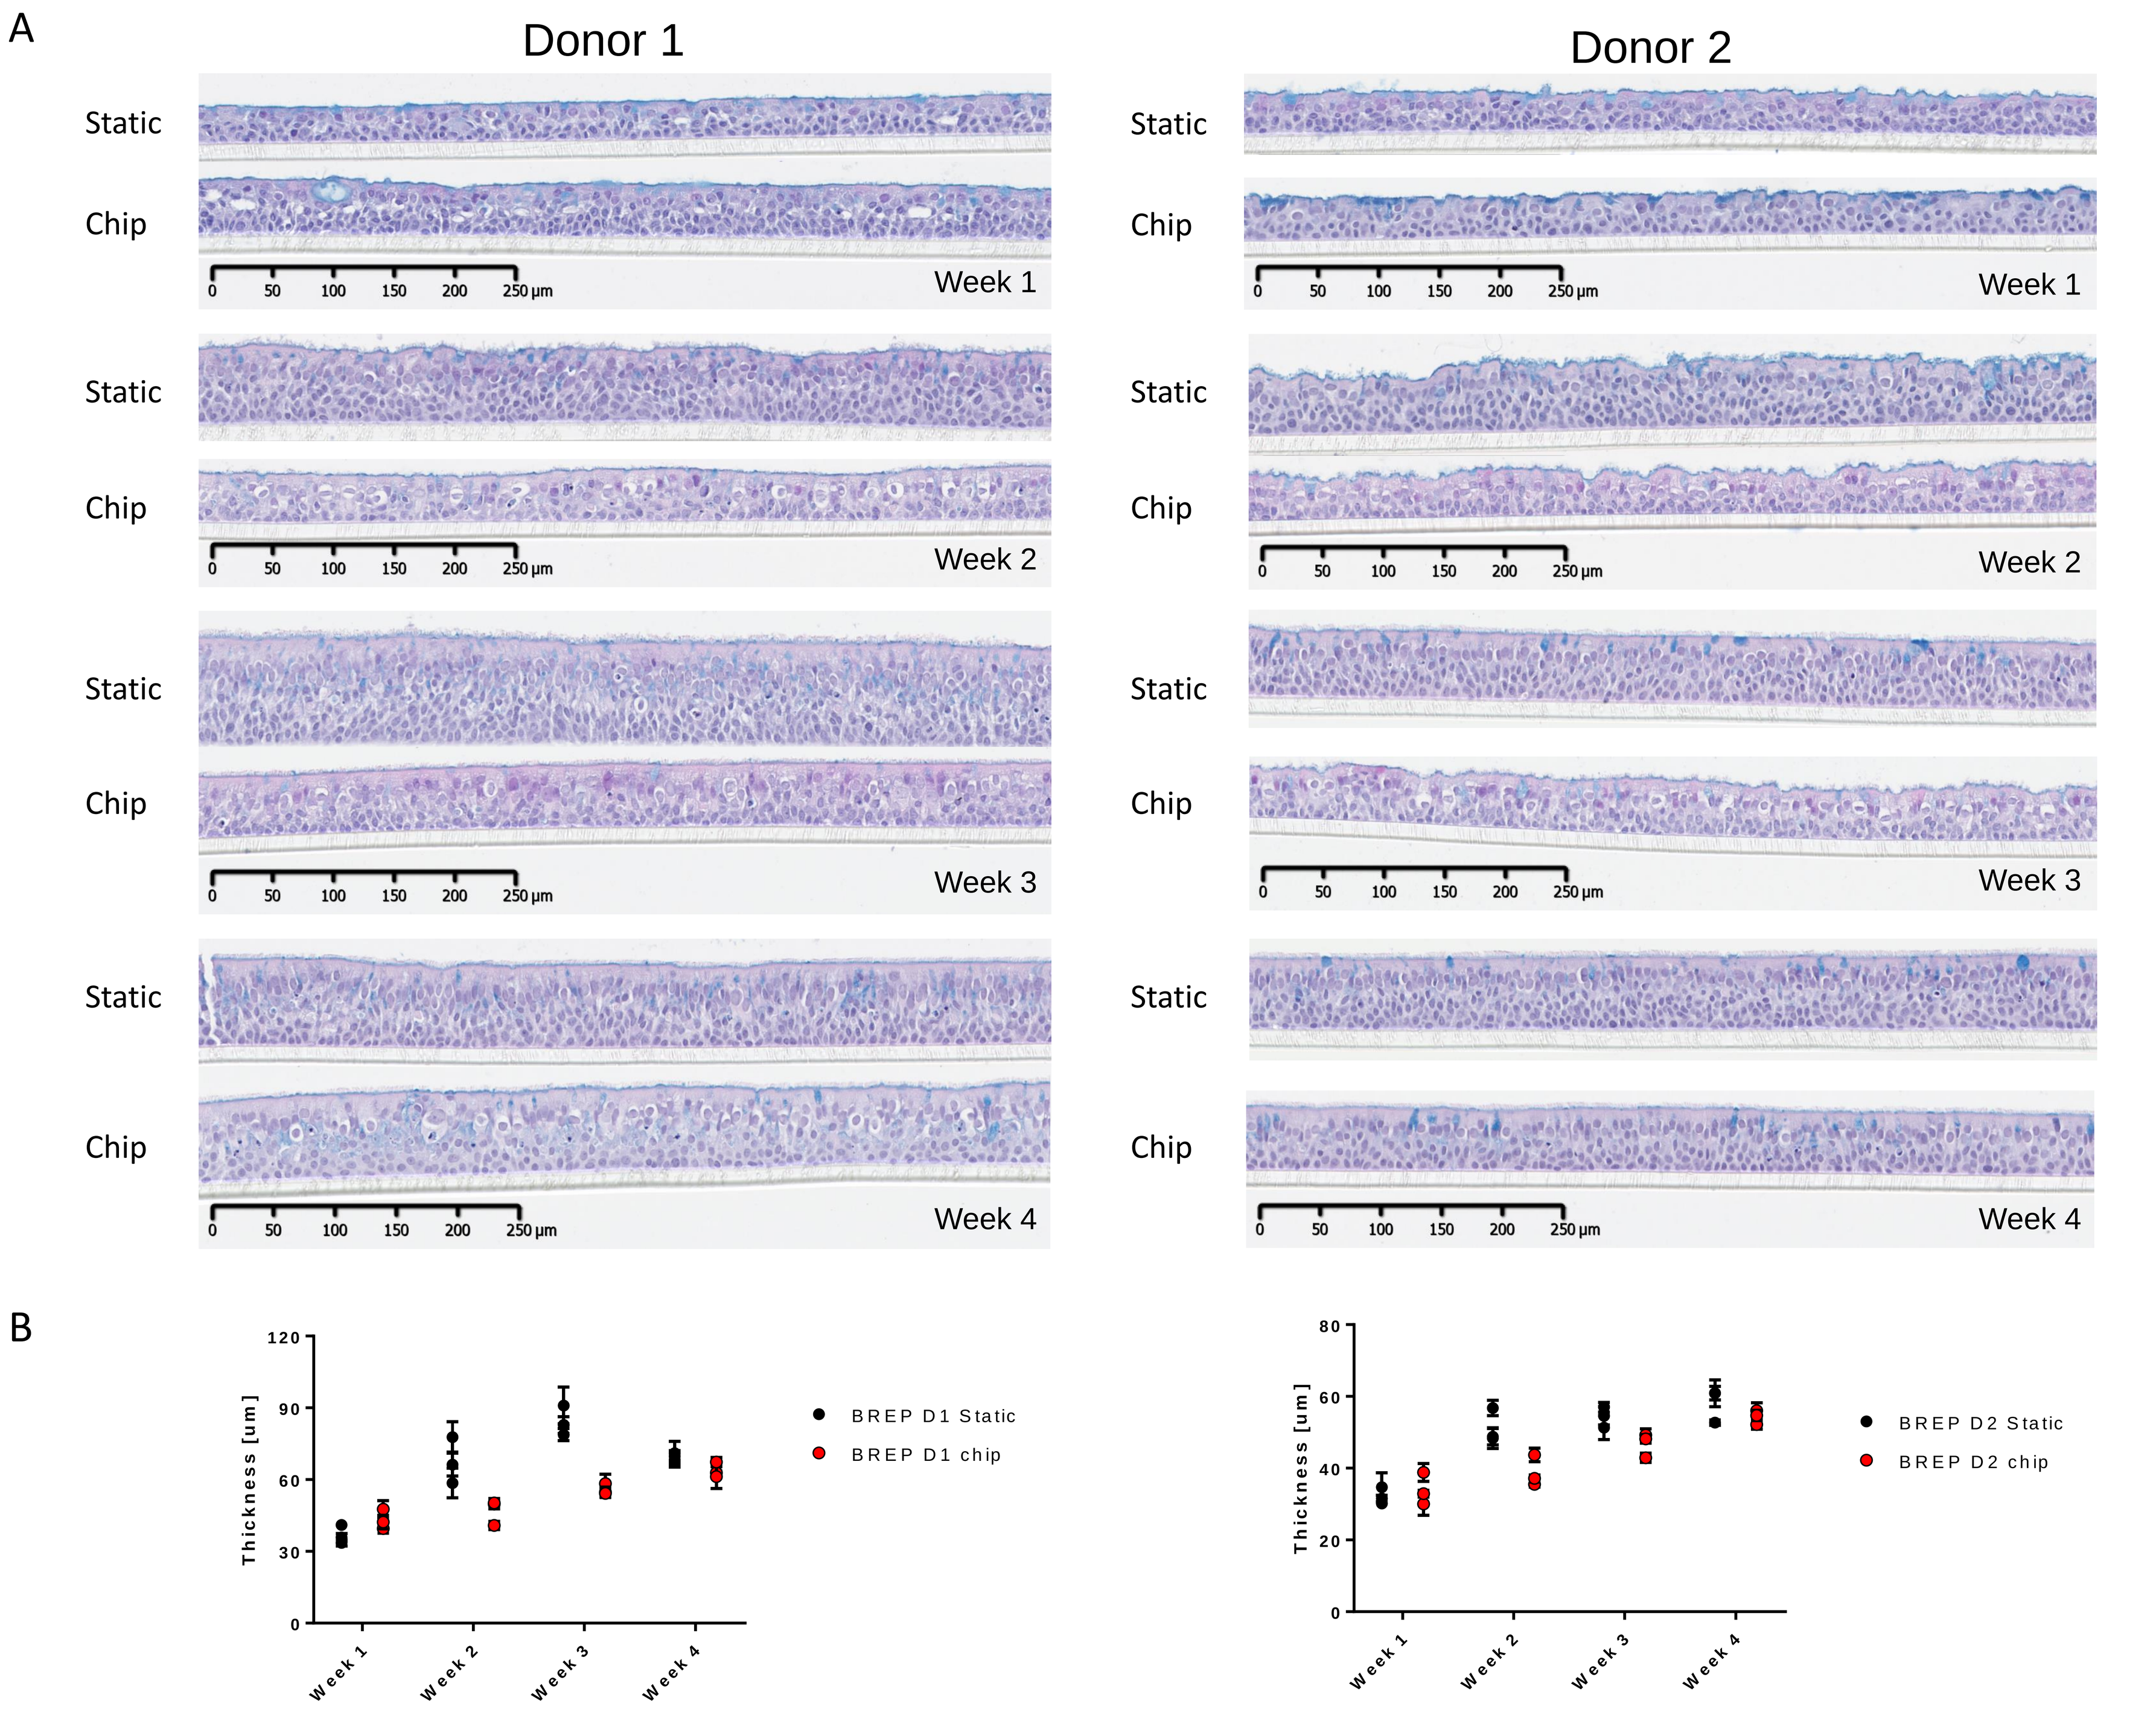
A
Donor 1
Donor 2
Static
Chip
Week 1
Week 2
Week 3
Week 4
0 50 100 150 200 250 µm
B
Thickness [um]
BREP D1 Static
BREP D1 chip
BREP D2 Static
BREP D2 chip

Figure 5: ***A)*** *Bronchial tissue morphology was evaluated every week following H&E/Alcian blue staining in both conditions. Objective 20x.* ***B)*** *Tissue thickness was evaluated by measuring the epithelium thickness in 3 different tissues (6 regions per tissue) every week. Each dot on the graphs shows the mean thickness ± standard deviation.*

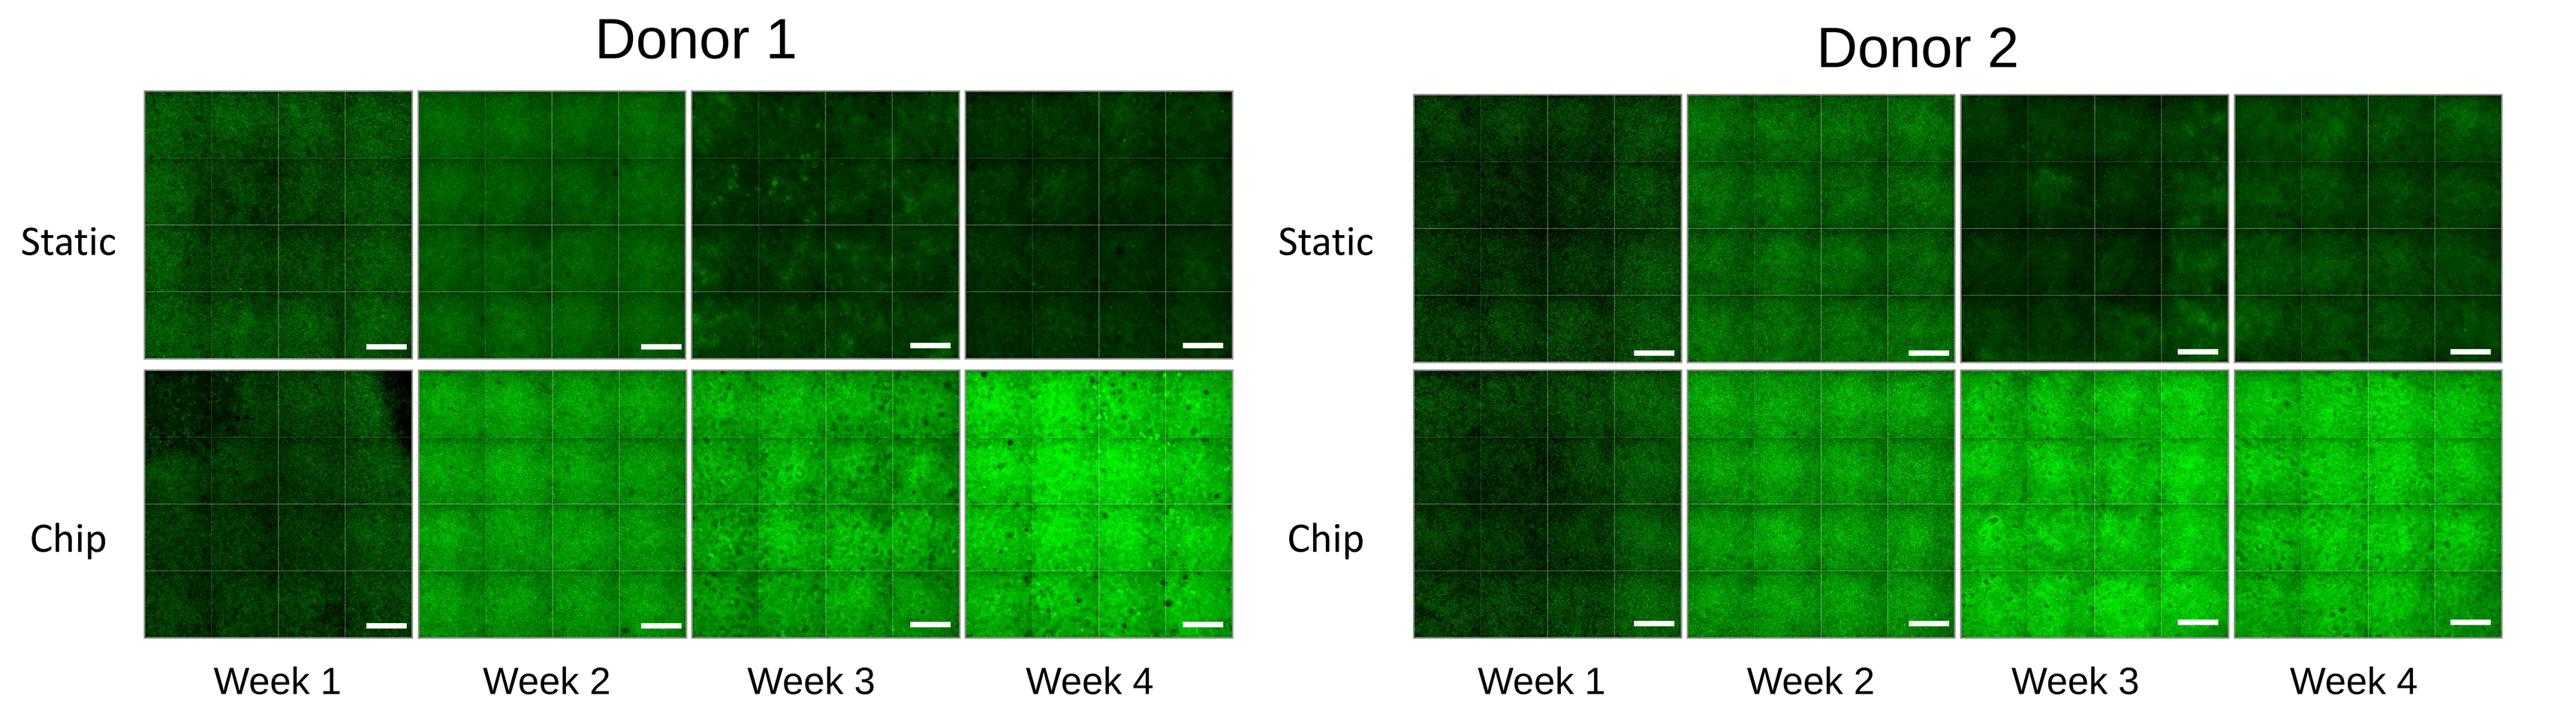
Donor 1
Donor 2
Static
Chip
Week 1
Week 2
Week 3
Week 4

Figure 6: *Tissues were stained every week with an anti β-tubulin 4 antibody to detect the cilia. All the tissues were stained and acquired using the same parameter. The acquired region shows the central region of the tissue and covers around ~60% of the entire surface. Objective: 10x. Scale bar corresponds to 200 µm.*

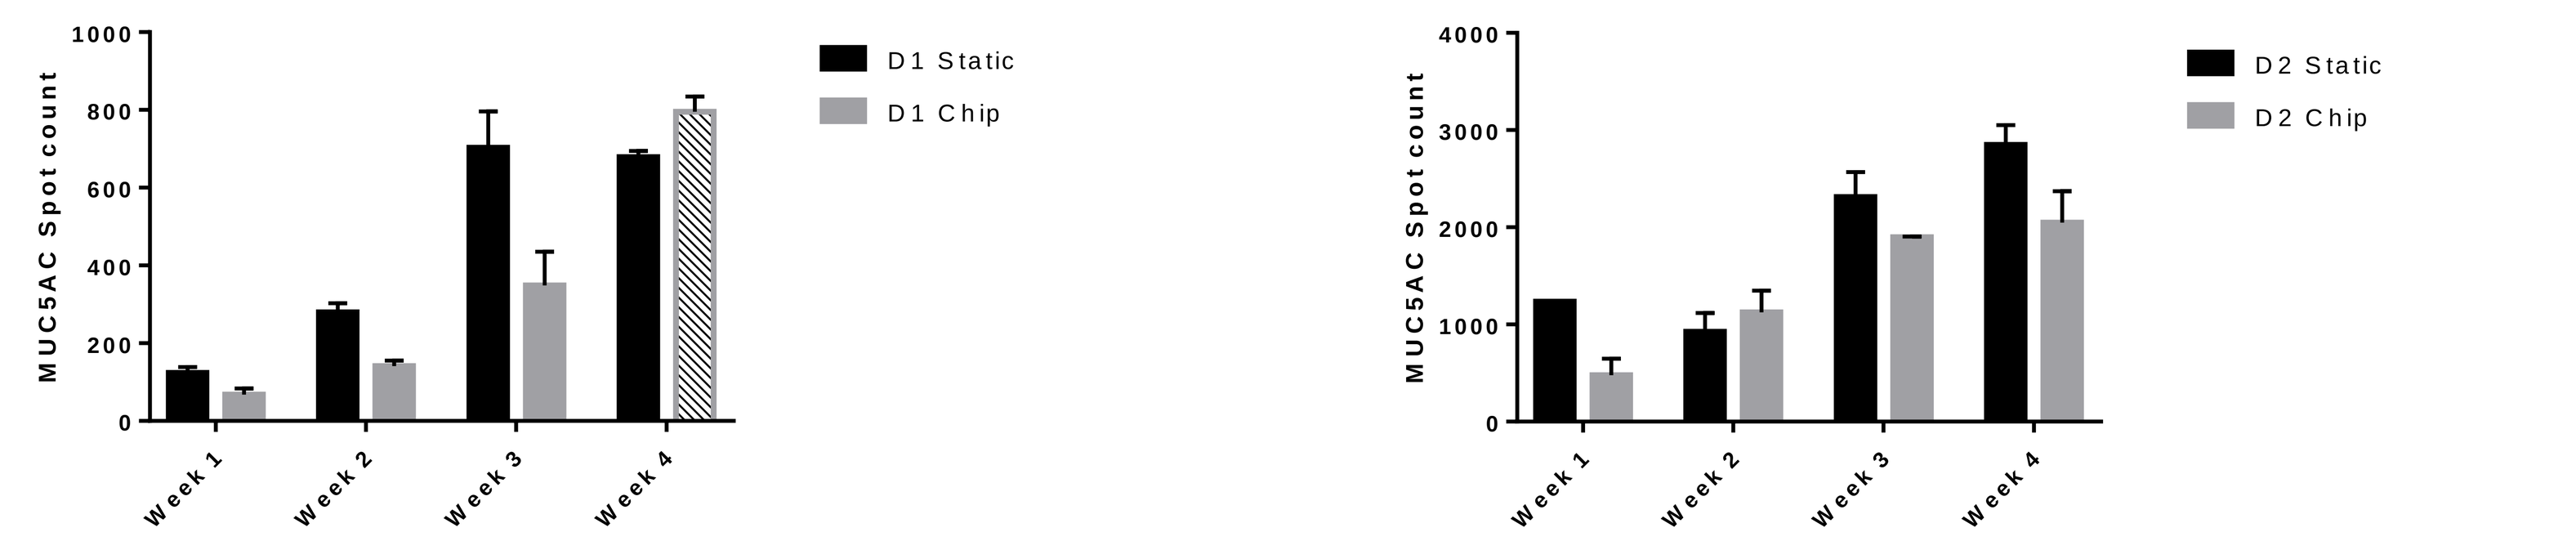
MUC5AC Spot count
D1 Static
D1 Chip
D2 Static
D2 Chip
Week 1
Week 2
Week 3
Week 4

Figure 7: *Tissues were stained every week with an anti mucin 5AC antibody to detect the goblet cells. All the tissues were stained and acquired using the same parameter. A central region corresponding to ~60% of the tissues' surface area was acquired, and the number of goblet cells was counted using a CX-7 platform. The spot count performed on tissues originating from donor 1 at week 4 (dashed column) is not accurate, as a strong background made the identification of positive MUC5AC spots difficult. MUC5AC = Mucin 5AC; D = Donor.*

Conclusion & discussions

The chip platform used in this study is an adaptation of a previously tested platform that successfully demonstrated crosstalk between bronchial and liver tissues (Bovard et al. 2018). Here we showed that the current procedure used to mature bronchial tissues at the ALI could be further improved using the chip technology.
We observed that :

- The tissues matured in the chip were thinner and showed a better polarization of the ciliated cells and a denser ciliation than tissues matured in static conditions. Tissues in standard conditions were composed of several cell layers between the basal and apical layer of cells, a phenomenon less observed in tissues matured in the chip.
- Tissues matured in the chip showed a denser or accelerated ciliation from Week 2 to Week 4 compared to tissues grown in standard conditions.
- The maturation of the tissues within the chip did not affect their functionality. TEER and CYP1A1/1B1 measures were similar to static cultures. The CBF was up to 15% lower in tissues matured in the chip.

The presence of a continuous medium flow—suppressing potential microenvironments in the culture medium below the tissues and causing shear stress—associated with a decreased supply of growth factor (lower medium change regimen) improved and accelerated ciliated cell differentiation and maturation. The thinner tissue epithelium might have to do with better nutrient supply and therefore a lower tendency for the basal cells to become hyperplastic. Maturation of tissue with medium flow within the chip not only improved bronchial tissue characteristics but also decreased the frequency of medium change by half to obtain mature and functional tissues.

Competing Financial Interest – The research described in this poster was sponsored by Philip Morris International