MOLECULAR MARKERS OF EARLY STAGE COPD IN SERUM, SPUTUM, AND NASAL EPITHELIUM FROM A CASE-CONTROL STUDY

Nveed Chaudhary (in alphabetical order), Ashraf Elamin, Emmanuel Guedj, Julia Hoeng, Nikolai Ivanov, Karsta Luettich, Patrice Leroy, Florian Martin, Manuel C. Peitsch, Thomas Schneider, Alain Sewer, Marja Talikka, Bjoern Titz, and Gregory Vuillaume

Philip Morris International R&D, Philip Morris Products S.A., Quai Jeanrenaud 5, 2000 Neuchâtel, Switzerland

This work was funded by Philip Morris International

Introduction

COPD is a multicomponent disease of emphysema, chronic bronchitis, and small airway obstruction mainly caused by long-term exposure to tobacco smoking. COPD takes an average 20-25 years to develop, and while its key pathological stages are relatively well described, its onset is as of yet poorly characterized. In addition, its considerable heterogeneity has hampered the identification of biomarkers that could be leveraged in diagnosis, patient stratification, treatment or risk prediction of disease development.

Unlike the other clinical studies on COPD, our COPD Biomarker Id Study focused on the early stage disease and smokers who do not yet show reduced FEV1 measurement.
This work describes a molecular level (proteomics, transcriptomics, lipidomics) assessment of various biological samples obtained from a clinical study with the primary objective to identify a biomarker or panel of biomarkers for the differentiation of subjects with COPD, current smokers, former smokers and never-smokers.

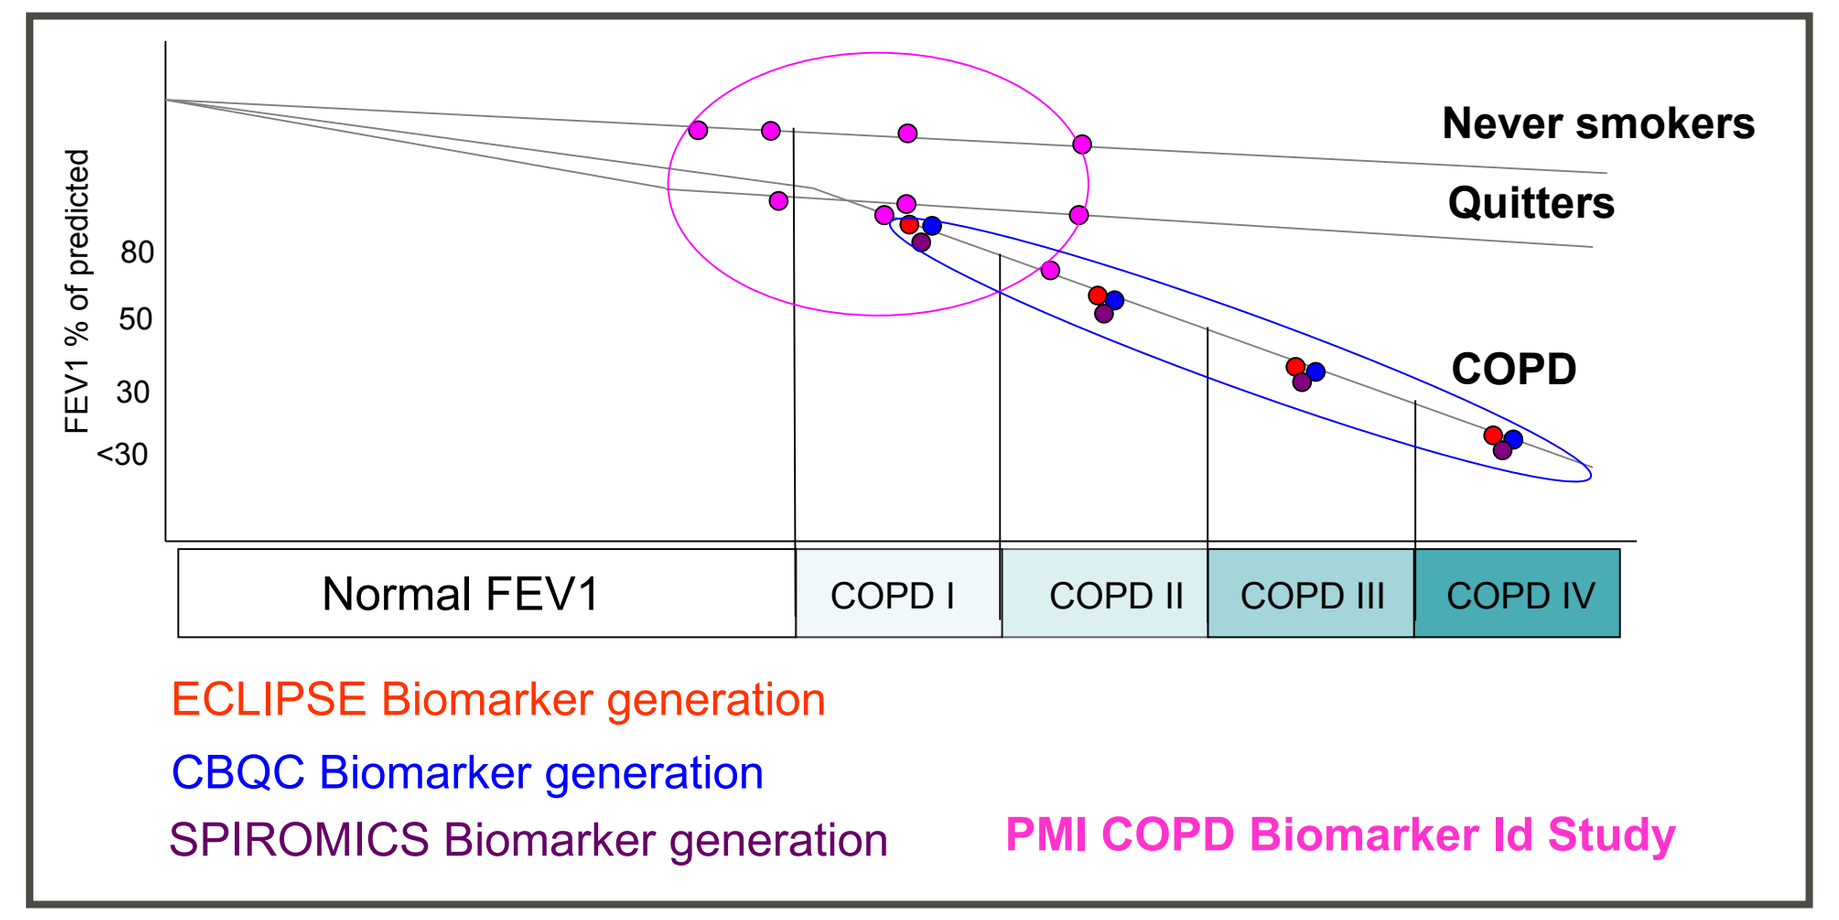
Never smokers
Quitters
COPD
FEV1 % of predicted
80
50
30
<30
Normal FEV1
COPD I
COPD II
COPD III
COPD IV
ECLIPSE Biomarker generation
CBQC Biomarker generation
SPIROMICS Biomarker generation
PMI COPD Biomarker Id Study

Study Description

We conducted a non-interventional, observational case-control design study in the United Kingdom, approved by a UK National Health Service (NHS) Ethics Committee (The Black County Ethics Committee) and in compliance with Good Clinical Practice guidelines. The study has been registered on ClinicalTrials.gov with identifier NCT01780298.

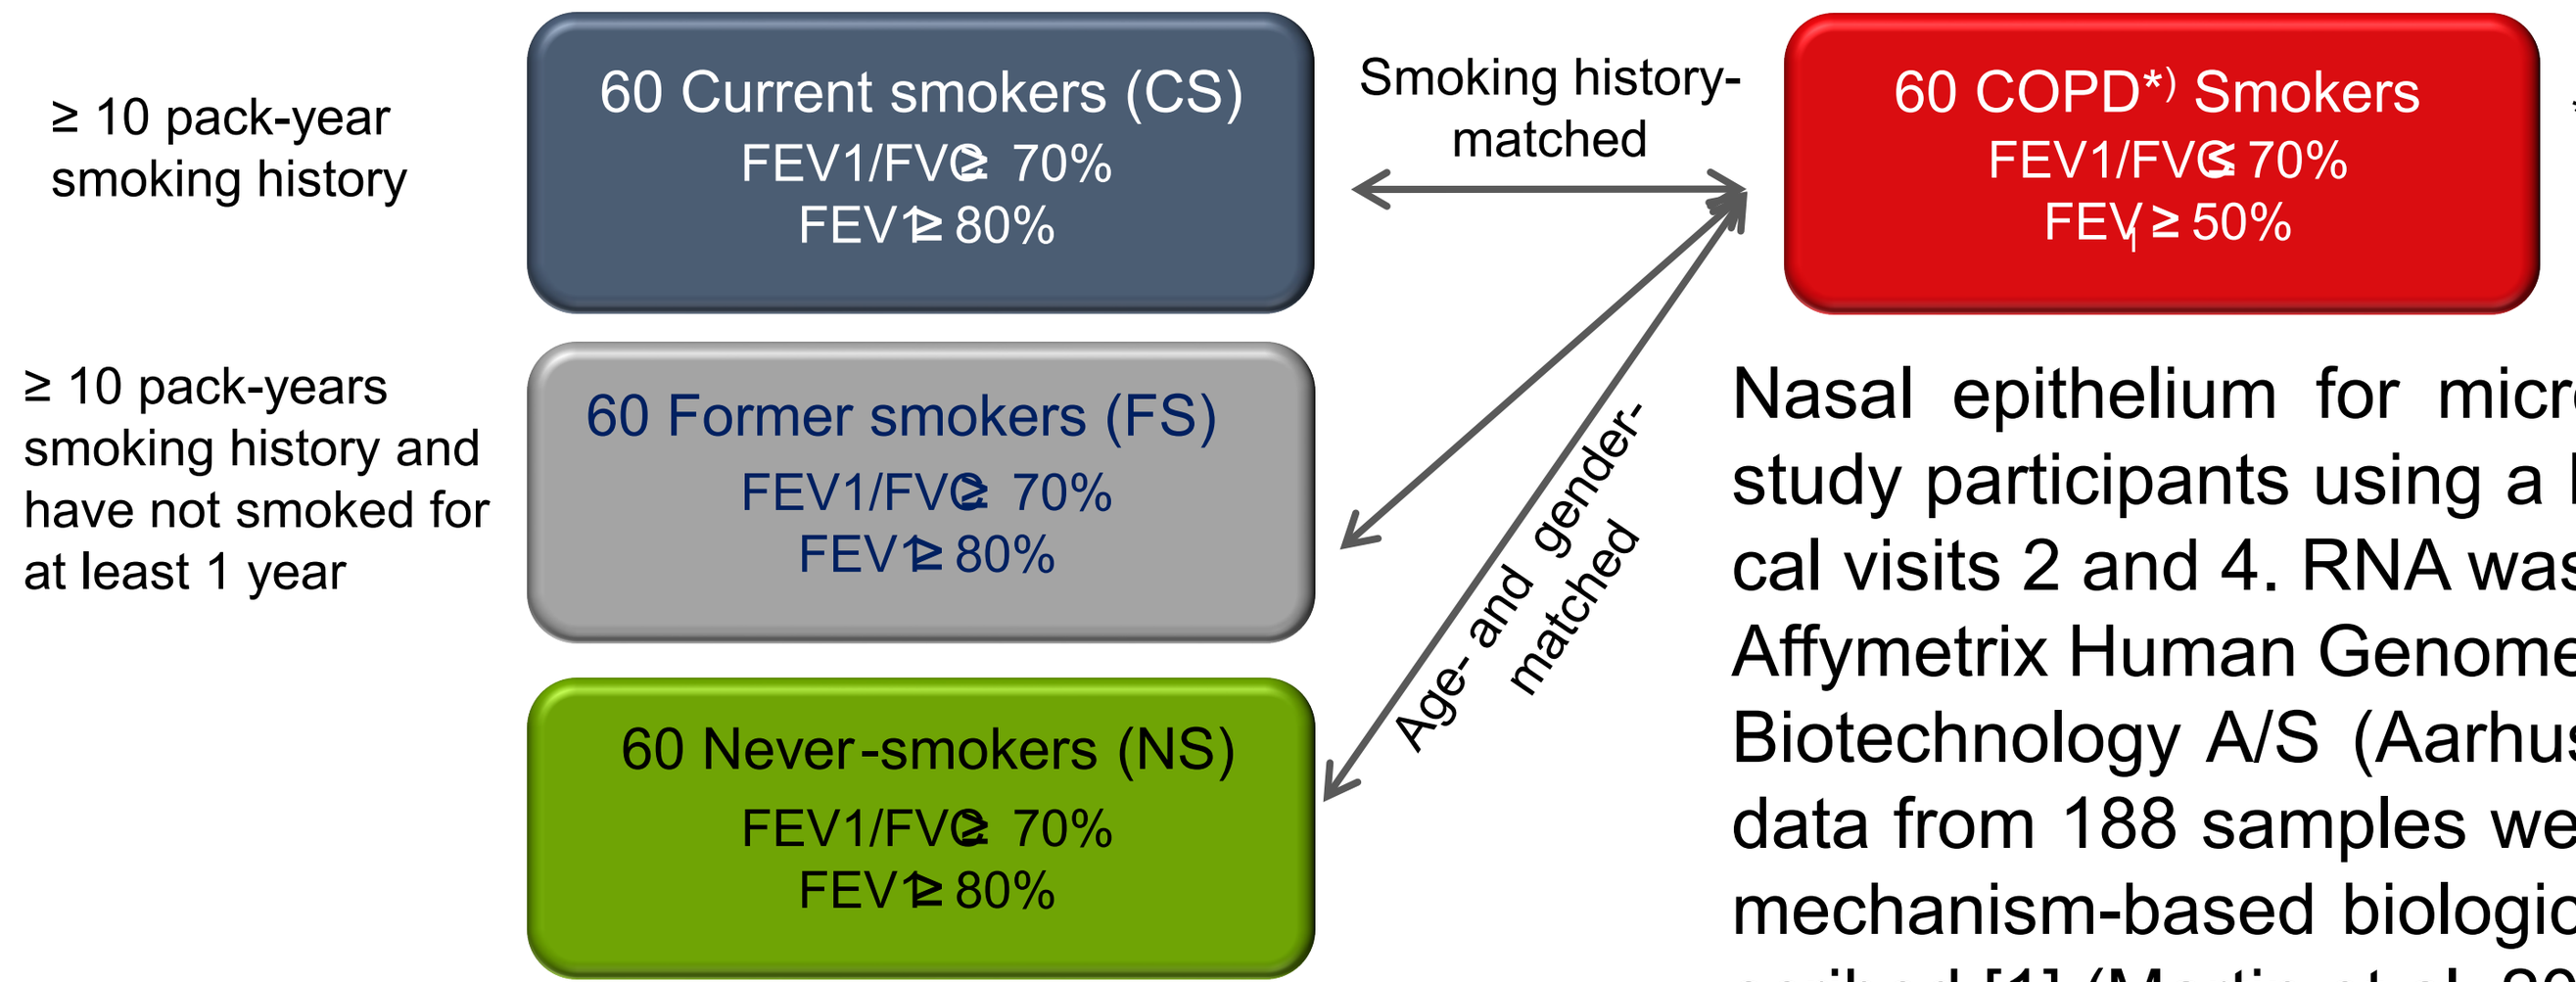
≥ 10 pack-year smoking history
60 Current smokers (CS)
FEV1/FVC ≥ 70%
FEV1 ≥ 80%
Smoking history-matched
60 COPD*) Smokers
FEV1/FVC ≤ 70%
FEV1 ≥ 50%
*) Stage I and IIa based on GOLD 2009 guidelines
≥ 10 pack-years smoking history and have not smoked for at least 1 year
60 Former smokers (FS)
FEV1/FVC ≥ 70%
FEV1 ≥ 80%
60 Never-smokers (NS)
FEV1/FVC ≥ 70%
FEV1 ≥ 80%
Age- and gender-matched

Nasal epithelium for microarray analysis was collected from all study participants using a Rhinoprobe® plastic curette during clinical visits 2 and 4. RNA was extracted, processed and hybridized to Affymetrix Human Genome U133 Plus 2.0 Arrays by AROS Applied Biotechnology A/S (Aarhus, Denmark). Following QC, microarray data from 188 samples were available for analysis by quantitative, mechanism-based biological network algorithms as previously described [1] (Martin et al. 2014).

Induced sputum was collected from all study participants by inhalation of hypertonic saline solution at visit. Sputum samples were placed directly on ice and processed within 2 hours. Sputum plugs were selected and solubilized in dithiothreitol (DTT), and the cell pellet was collected by centrifugation for cytometry evaluation, while the supernatant was collected for subsequent proteomic analysis. Non-targeted proteomics was performed by LC-MS/MS employing isobaric tags using Tandem Mass Tag™ 6-plex (TMTsixplex™) Reagents (Thermo Scientific). Spectrometric data were compared against the human reference proteome, and Proteome Discoverer™ software was employed for peptide-to-protein assignment. Peptide-level data were then further analyzed in the R statistical environment. Each TMT™ reporter ion set was normalized to its median, and protein expression values were calculated as the median of these normalized peptide-level quantitation values [2] (Herbrich et al, 2012). Only proteins quantified for at least 2/3 of the samples of each study group were considered.
For blood-based biomarker analysis, blood was collected in 8.5 mL serum-separating tubes (SST) during visit 2, allowed to clot at room temperature for 30 min and centrifuged. Serum samples were aliquotted at stored at -80°C until further use. Analyses of lipid species were performed in serum samples from a subset of study subjects including 40 never-smokers, 40 former smokers, 40 smokers, and 40 COPD smokers by Zora Biosciences Oy (Espoo, Finland) using MS-based methodologies. Mixed linear models were employed to identify significant differences in serum levels of individual lipid species between study groups. Study subjects' age, BMI, alcohol intake and intake of lipid-modifying drugs (e.g. statins) were taken into consideration as a potential confounders and/or covariates.

Nasal Transcriptomics

Biological network analysis indicates that cigarette smoking significantly impacts cell stress, cell fate and inflammatory processes (CS vs NS). While most of the affected mechanisms are less impacted in former smokers, perturbations persist in processes related to DNA damage (FS vs NS).

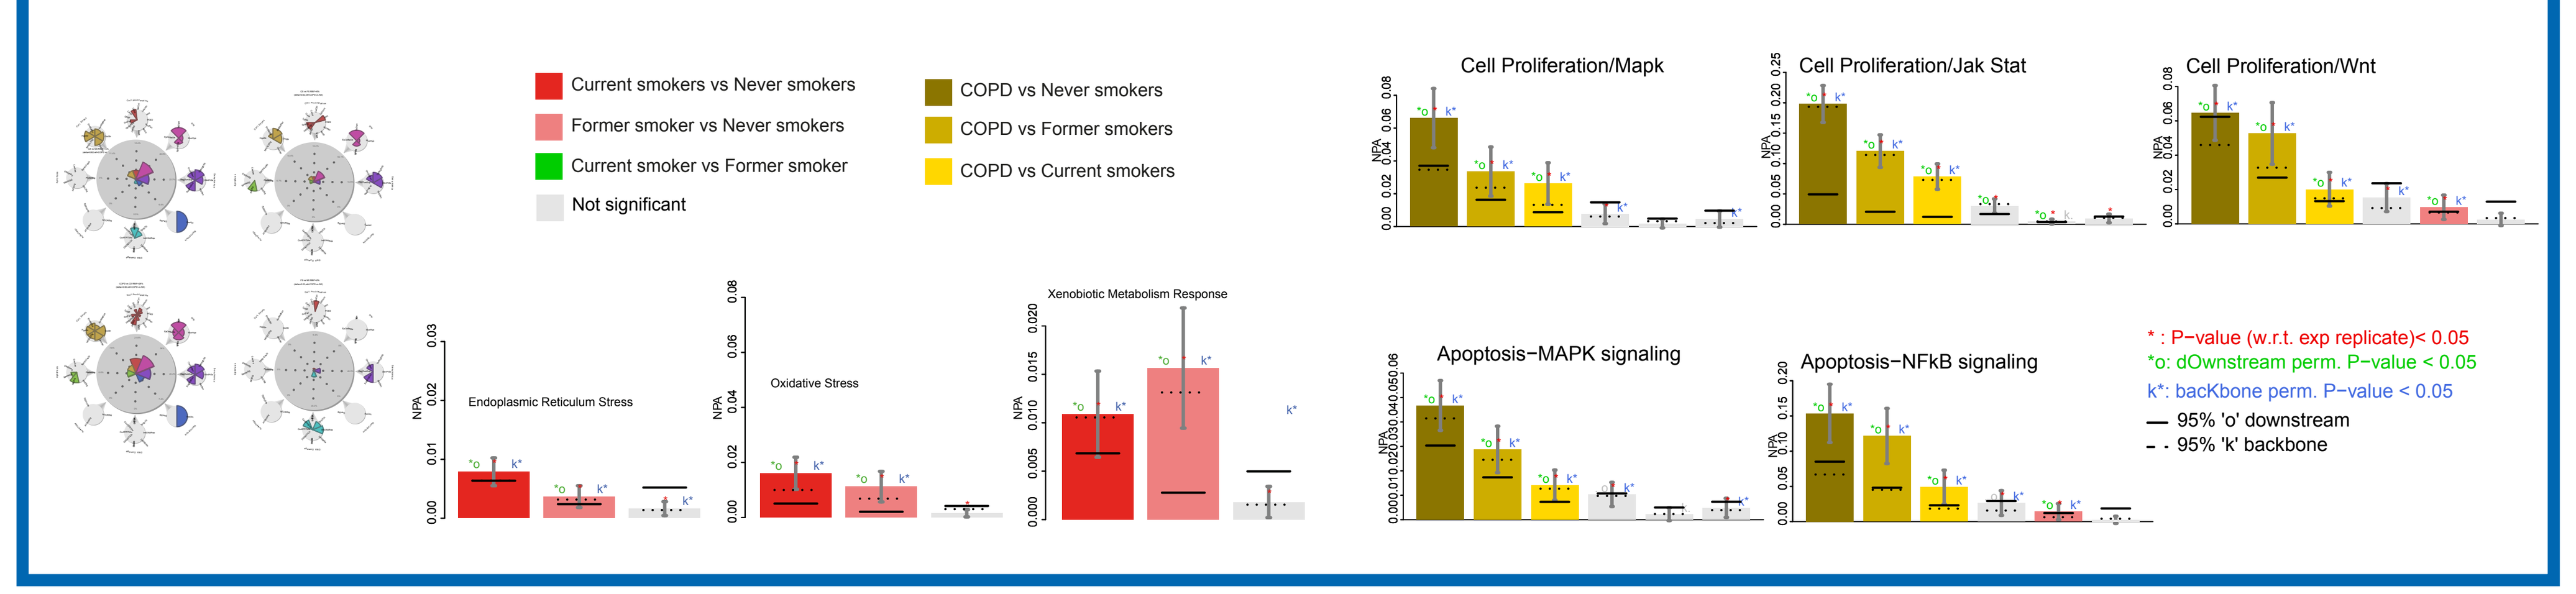
Current smokers vs Never smokers
Former smoker vs Never smokers
Current smoker vs Former smoker
Not significant
COPD vs Never smokers
COPD vs Former smokers
COPD vs Current smokers
Cell Proliferation/Mapk
Cell Proliferation/Jak Stat
Cell Proliferation/Wnt
Endoplasmic Reticulum Stress
Oxidative Stress
Xenobiotic Metabolism Response
Apoptosis−MAPK signaling
Apoptosis−NFkB signaling
* : P−value (w.r.t. exp replicate)< 0.05
*o: dOwnstream perm. P−value < 0.05
k*: bacKbone perm. P−value < 0.05
95% 'o' downstream
95% 'k' backbone
NPA

Sputum Proteomics

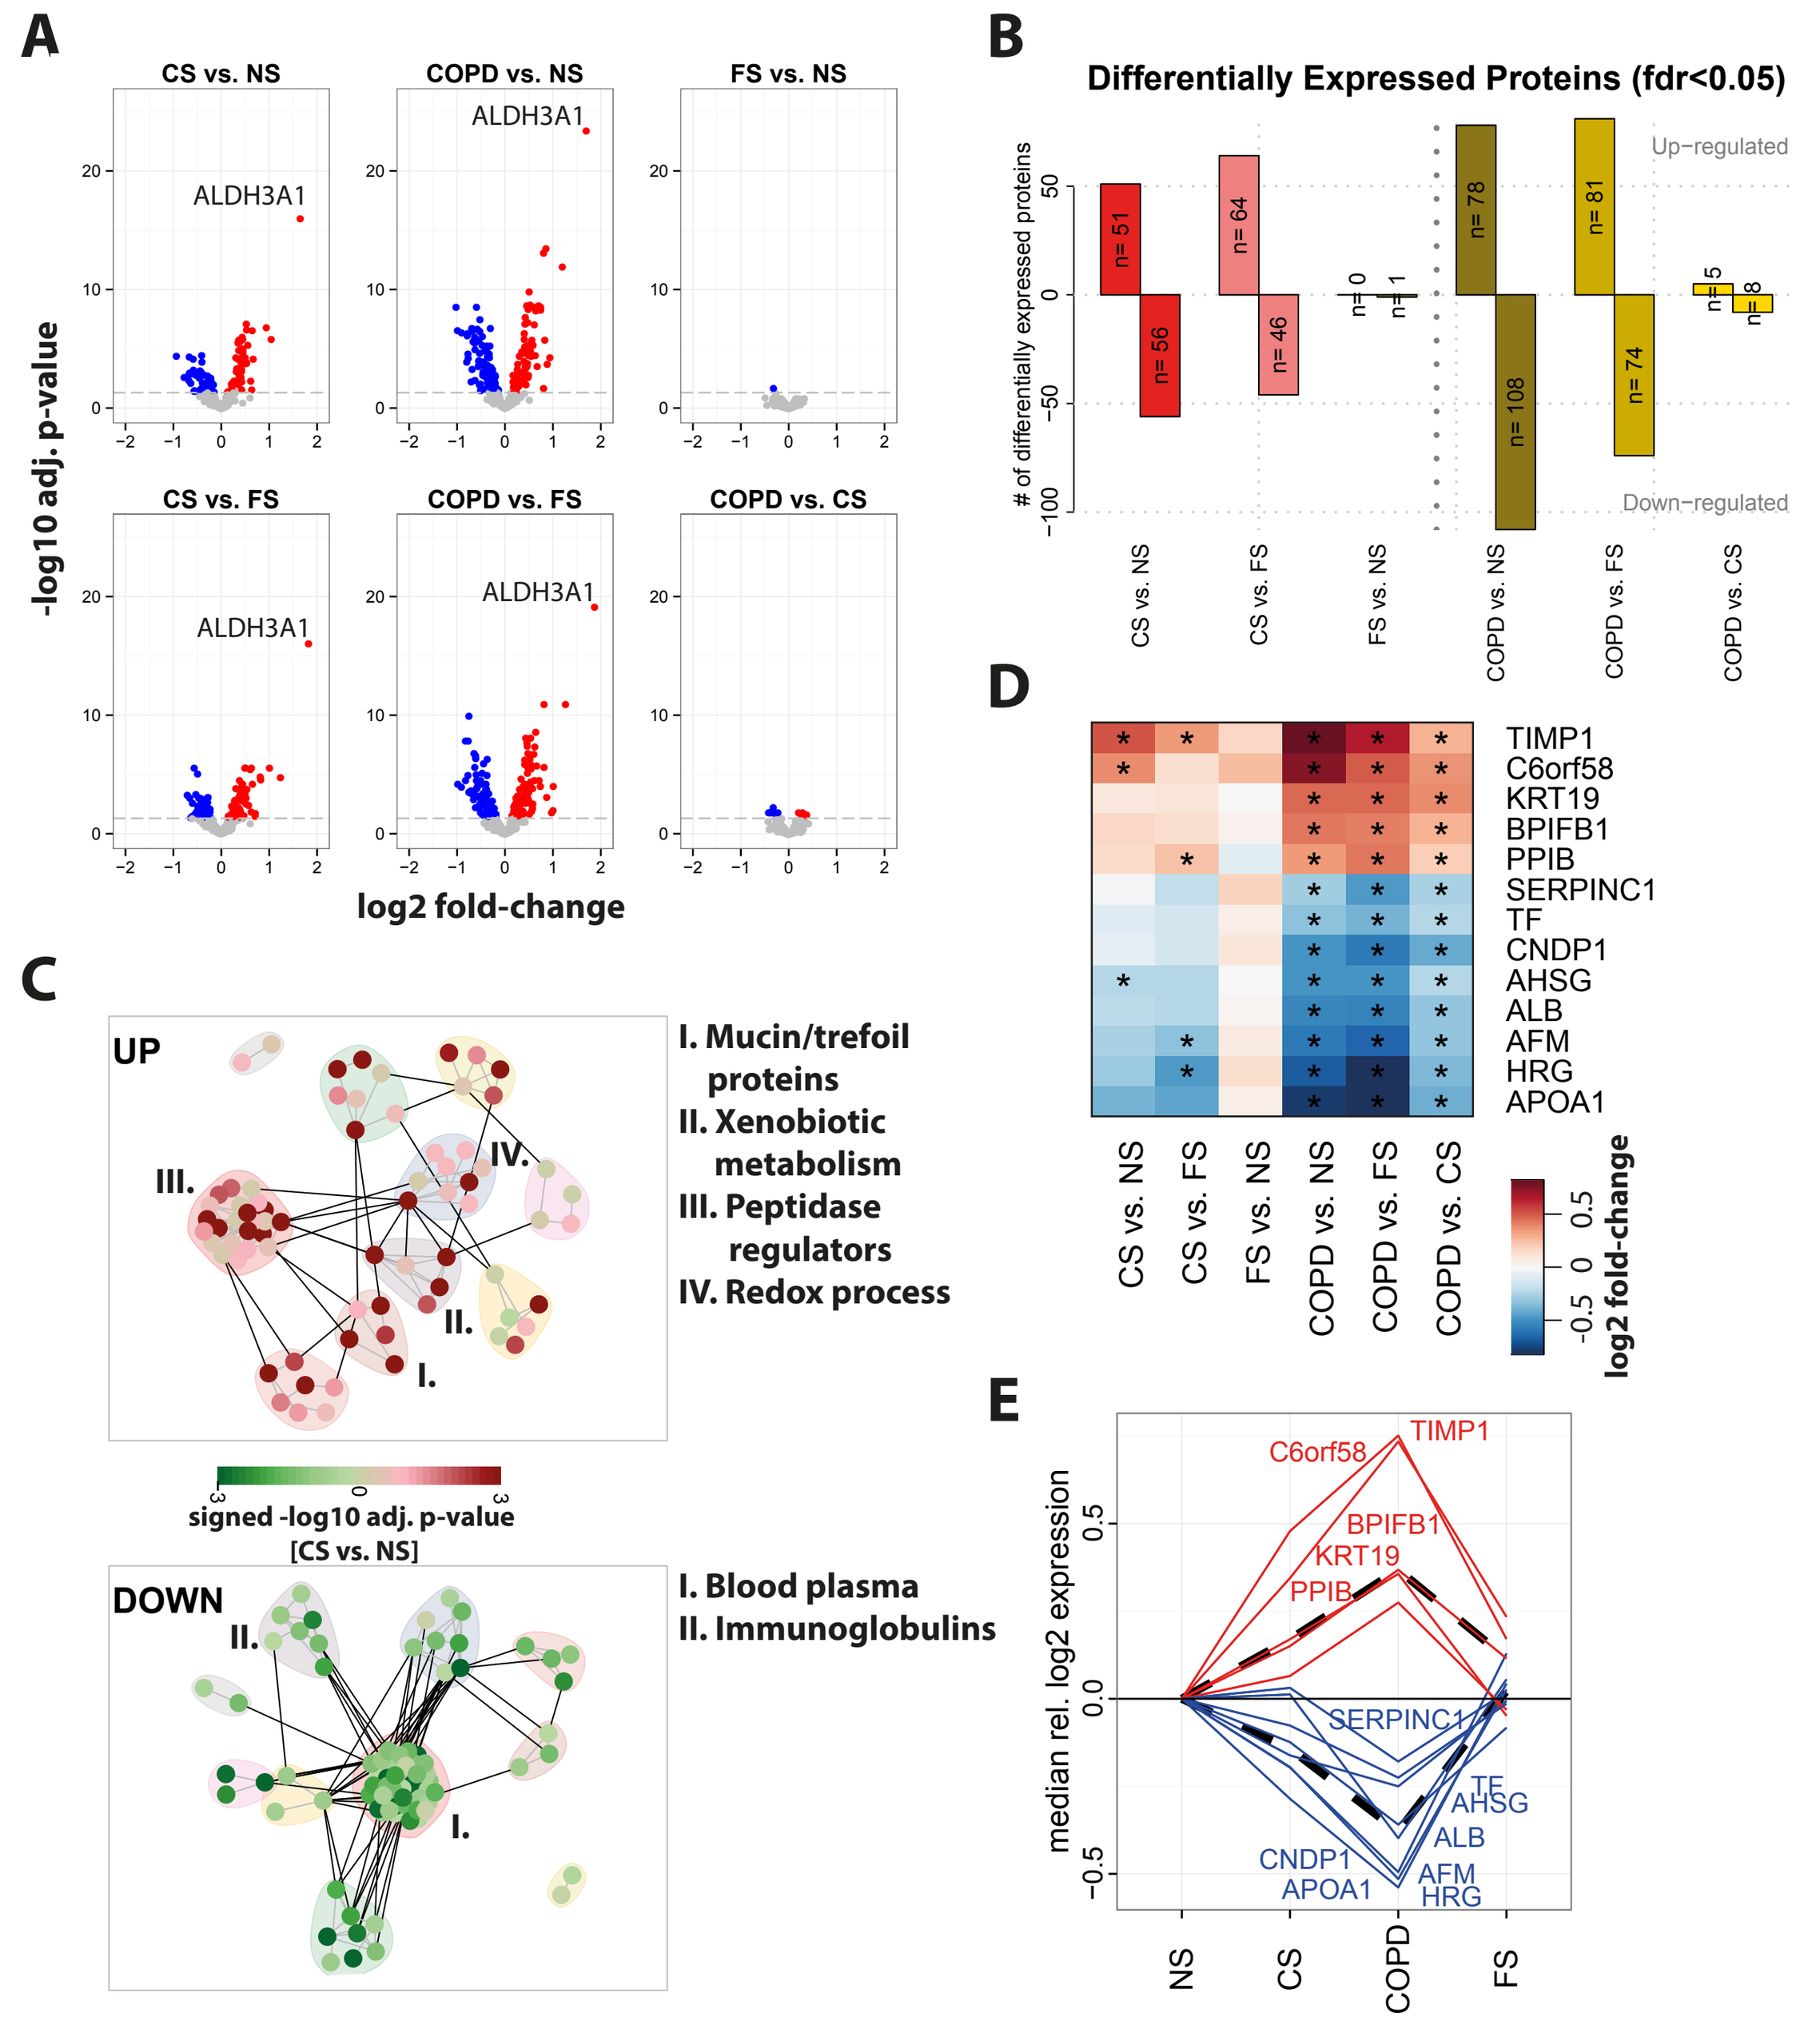
A
CS vs. NS
COPD vs. NS
FS vs. NS
CS vs. FS
COPD vs. FS
COPD vs. CS
ALDH3A1
-log10 adj. p-value
log2 fold-change
B
Differentially Expressed Proteins (fdr<0.05)
Up−regulated
Down−regulated
of differentially expressed proteins
n= 51
n= 56
n= 64
n= 46
n= 0
n= 1
n= 78
n= 108
n= 81
n= 74
n= 5
n= 8
C
UP
I. Mucin/trefoil proteins
II. Xenobiotic metabolism
III. Peptidase regulators
IV. Redox process
signed -log10 adj. p-value [CS vs. NS]
DOWN
I. Blood plasma
II. Immunoglobulins
D
TIMP1
C6orf58
KRT19
BPIFB1
PPIB
SERPINC1
TF
CNDP1
AHSG
ALB
AFM
HRG
APOA1
log2 fold-change
E
median rel. log2 expression
NS
CS
COPD
FS

107 proteins were found to be differentially expressed in sputum of current vs. never-smokers, and 13 proteins showed differential abundance in the sputum of COPD smokers compared to healthy current smokers (FDR adjusted p-value < 0.05) (**A**,**B**).
Functional association-based analysis for current vs. never-smokers indicates clusters of mucins/trefoil proteins, proteins involved in the xenobiotic and oxidative stress response, and peptidases and peptidase regulators among the over-abundant proteins and clusters of down-regulated antibodies and plasma proteins (**C**).
The 13 differentially expressed proteins in COPD patients vs. current smokers were also seen to be significantly differentially expressed when comparing COPD with former smokers, while no significant differences in sputum protein abundances were noted when comparing never- with former smokers (**D**). Interestingly, for most of these proteins differential expression appears to be already present in current smokers, and thus reflects an aggravation of the smoke exposure effects in COPD patients (**E**, dashed line shows median trend for up- and down-regulated protein group).

Serum Lipidomics

Cigarette smoking affects a number of lipid molecules including eicosanoids, ceramides and sphingomyelins, and some of these effects are irreversible upon smoking cessation. For example, 11,12-DHET and 14,15-DHET serum levels differ significantly between smokers and never-smokers, and smokers and former smokers, but not between former smokers and never-smokers. In contrast, 9-HODE serum levels are significantly different in smokers compared to never-smokers, while there are no significant differences in 9-HODE serum levels when comparing smokers with former smokers.

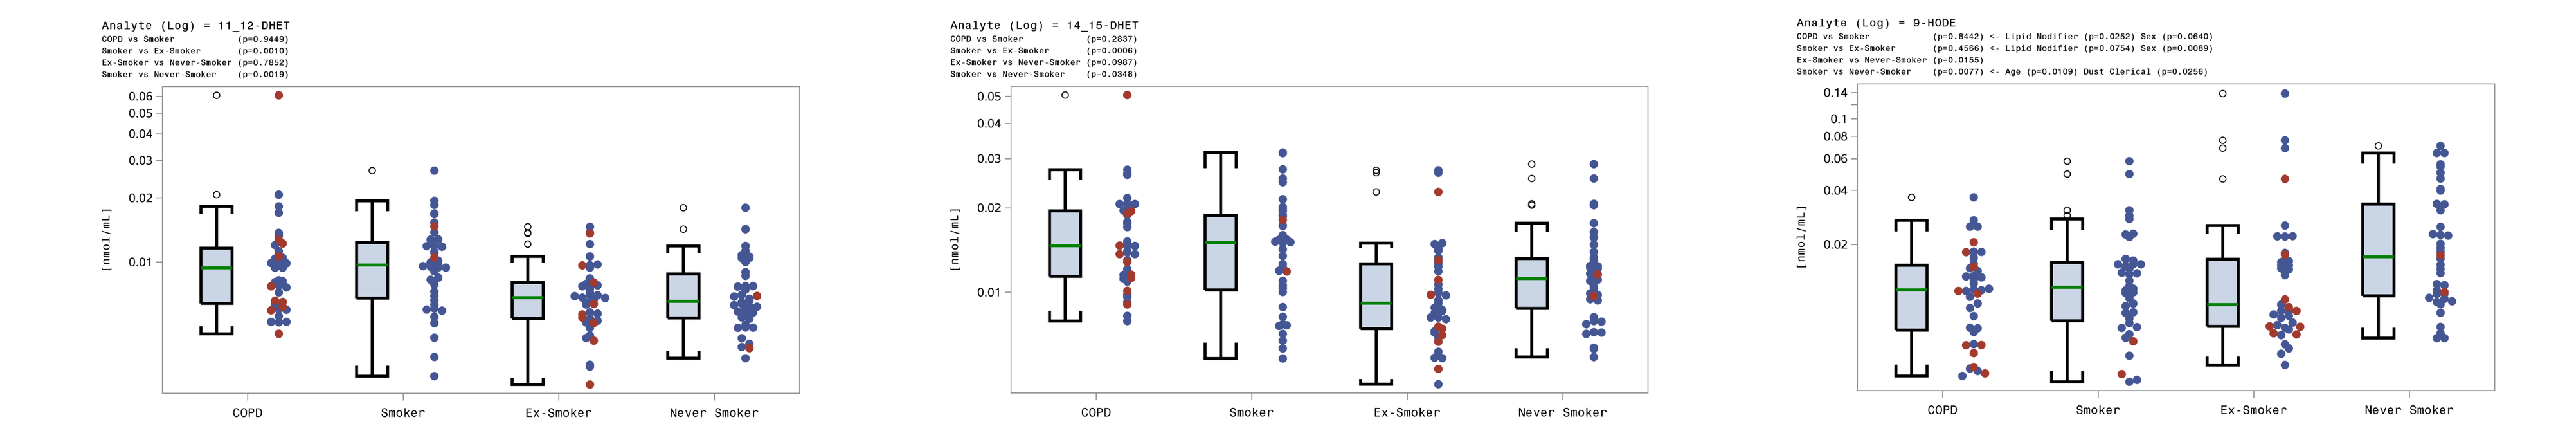
Analyte (Log) = 11_12-DHET
Analyte (Log) = 14_15-DHET
Analyte (Log) = 9-HODE
COPD
Smoker
Ex-Smoker
Never Smoker

The majority of lipid molecules examined are decreased in COPD patients compared to asymptomatic smokers suggesting that COPD-specific mechanisms exert specific effects on the serum lipid profile. For example, compared to smokers, COPD patients had significantly lower levels of serum sphingolipids SM(d18:0/16:0), SM(d18:1/18:0), SM(d18:1/24:0), and SM(d18:1/24:1), and phospholipids PC(16:0/18:2), PC(16:0/22:6), and PC(18:0/22:6).

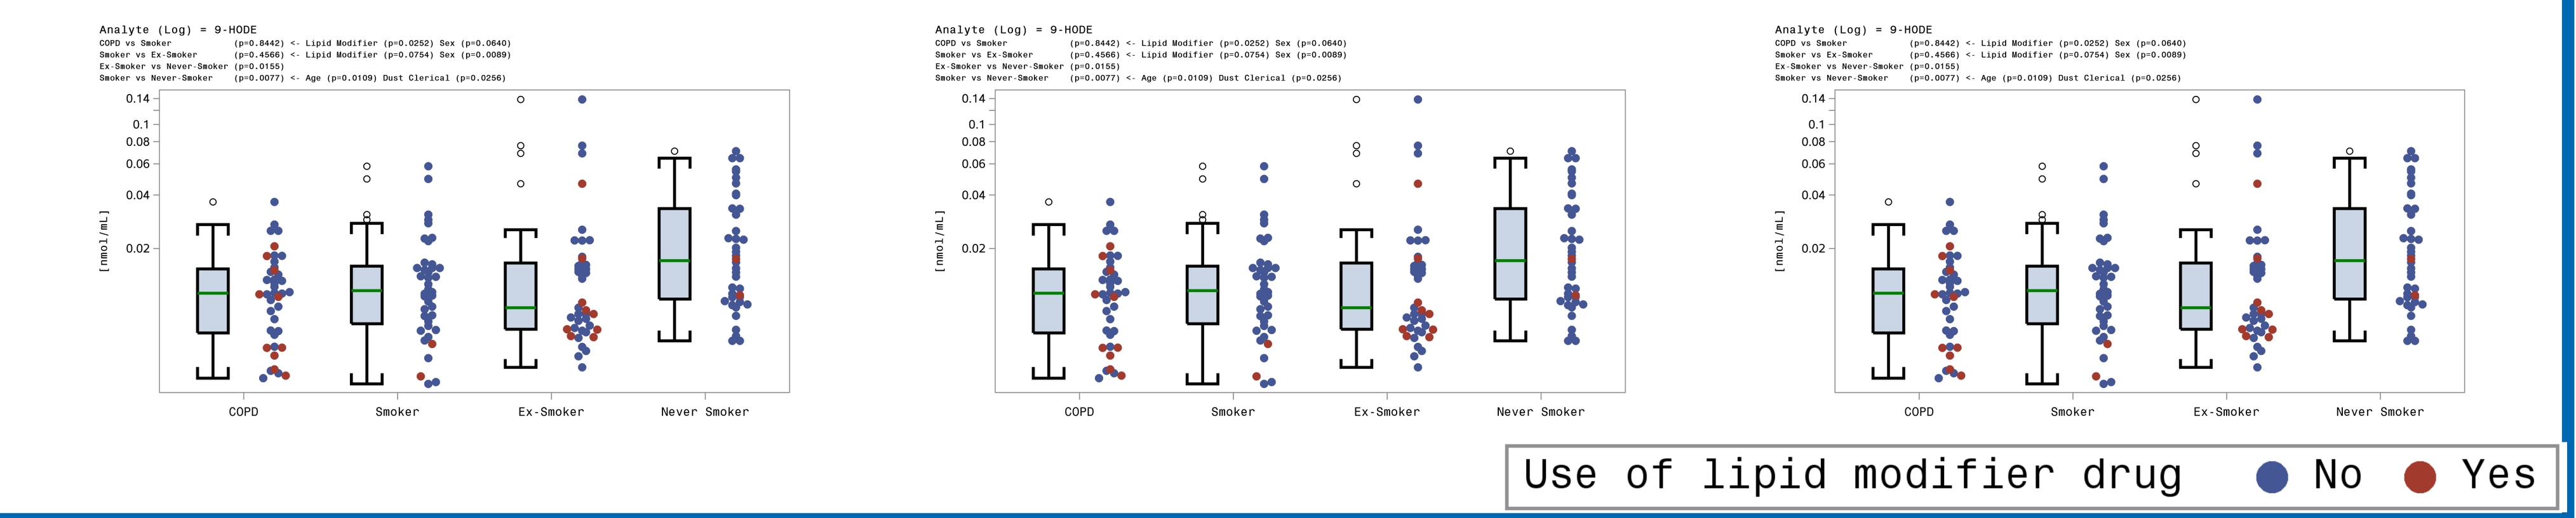
Analyte (Log) = 9-HODE
COPD
Smoker
Ex-Smoker
Never Smoker
Use of lipid modifier drug
No
Yes

Conclusions and Perspective

On the transcriptomic level, biological network analysis demonstrates the impact of cigarette smoking on several cellular processes incl. cell stress cell fate and inflammation which is at least partially reversible upon smoking cessation. Early stage disease development appears to further affect these processes.
Proteomics analysis shows that the abundance of sputum proteins linked to cellular stress responses and the protease-antiprotease balance is altered by cigarette smoking and that these alterations are "restored" upon smoking cessation, while they appear to be further aggravated by the development of early COPD.
Cigarette smoking also increases the serum levels of several bioactive, inflammatory lipids which are only partially restored when exposure ceases.
The affected RNA, protein and/or lipid molecules could be further explored as biomarkers with utility in product assessment studies.
Specific molecular changes observed in patients with early stages of disease could be further examined with respect to their potential to indicate disease risk.

References

[1] Martin F, Sewer A, Talikka M, Xiang Y, Hoeng J, Peitsch MC (2014) Quantification of biological network perturbations for mechanistic insight and diagnostics using two-layer causal models. BMC bioinformatics 15: 238
[2] Herbrich SM, Cole RN, West KP, Schulze K, Yager JD, Groopman JD, Christian P, Wu L, O'Meally RN, May DH, McIntosh MW, Ruczinski I (2012) Statistical Inference from Multiple iTRAQ Experiments without Using Common Reference Standards. Journal of Proteome Research 12: 594-604

PMI RESEARCH & DEVELOPMENT

2015 SRNT 21ST Annual Meeting

February 25-28, 2015
Philadelphia Marriott Hotel