Cigarette smoke dose-dependently exacerbates clinical symptoms and pathology in a mouse model of multiple sclerosis

K. Koshibu[†1], J. Ho[†2], W. Xia[2], K., L. Garcia[1], A. Kondylis[1], K. Luettich[1], B. Phillips[2], J. Hoeng[1], M. Peitsch[1]

[1]PMI R&D, Philip Morris Products S.A., Quai Jeanrenaud 5, CH-2000 Neuchâtel, Switzerland; [2]PMI R&D, Philip Morris International Research Laboratories Pte. Ltd., Science Park II, Singapore; † These authors contributed equally.

Introduction

Multiple sclerosis (MS) is an inflammatory autoimmune disease of the central nervous system. The etiology of the disease is complex and involves an interplay between genetic and environmental factors. Cigarette smoking has emerged as a major risk factor associated with the onset of MS[1, 2]. To further address this topic, we studied the effect of chronic cigarette smoke (CS) exposure in an experimental autoimmune encephalomyelitis (EAE) mouse model. Among several models of human MS, EAE is the most commonly employed animal model for investigating the pathogenesis of and therapeutic interventions for MS. In our study, we chose a myelin oligodendrocyte glycoprotein amino acid 35–55 (MOG35-35)-induced EAE mouse model to investigate the effect of CS exposure on EAE prevalence, onset, progression, severity, and spinal cord pathology. Our results show that CS can affect clinical symptoms and spinal cord lesions in a dose-dependent manner.

Methods

Animals: Adult C57BL/6 female mice were purchased from InVivos Pte. Ltd (Singapore). All animal experiments were approved by the International Animal Care and Use Committee, Association for Assessment and Accreditation of Laboratory Animal Care International, and National Advisory Committee for Laboratory Animal Research.

Exposure: Mice (~10 weeks old) were exposed to fresh air or mainstream smoke from 3R4F reference cigarettes (University of Kentucky) in whole-body exposure chambers. CS was diluted with filtered, conditioned air to three different target concentrations of total particulate matter (TPM): 150 µg/L (Low), 350 µg/L (Med), and 600 µg/L (High). The animals were gradually adapted to CS exposure over the course of 1 week and exposed to CS at the target TPM concentrations for another week prior to EAE induction. To avoid carbon monoxide toxicity, the daily exposure regimen was divided into four blocks of 1-h exposures, each of which was followed by a 0.5- or 1-h fresh air breaks. After the EAE induction, the exposure regimen was resumed for an additional 4 weeks. Uptake of CS was confirmed by analyses of urinary nicotine and nicotine metabolites. Urine was collected from a subset of animals on study days 9 and 12 over a 24-h period, starting from the beginning of exposure.

A

Exposure	Chamber	TPM (µg/L) ± SD	CO (ppm) ± SD	Nicotine (µg/L) ± SD
Fresh air	1	-5.4 ± 1.7 (n = 36)	0.1 ± 0.1 (n = 36)	0.1 ± 0.0 (n = 8)
3R4F Low	2	145.7 ± 4.0 (n = 36)	192.1 ± 7.3 (n = 36)	8.9 ± 0.7 (n = 21)
3R4F Med	3	346.1 ± 12.5 (n = 36)	359.6 ± 24.7 (n = 36)	22.6 ± 1.5 (n = 20)
3R4F High	4	576 ± 17.9 (n = 36)	574.4 ± 44.5 (n = 36)	39.9 ± 3.0 (n = 20)

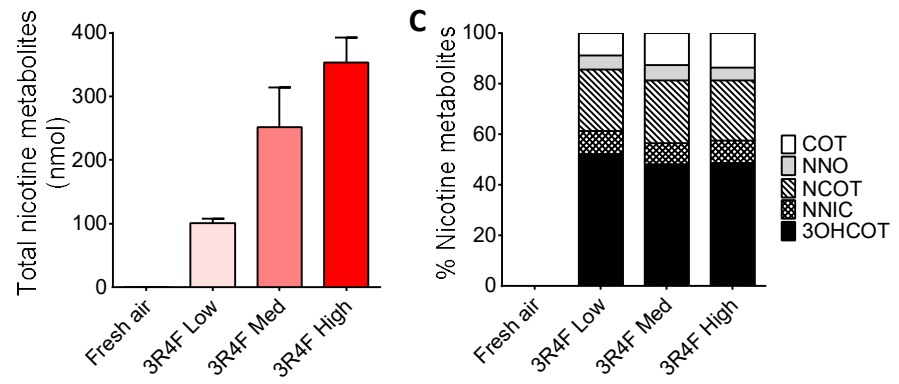
B
Total nicotine metabolites (nmol)
Fresh air
3R4F Low
3R4F Med
3R4F High
C
% Nicotine metabolites
COT
NNO
NCOT
NNIC
3OHCOT

Figure 1. CS and biomarkers of exposure
A) Average concentrations of TPM, CO, and nicotine in CS. B) Nicotine metabolite concentrations. C) Proportions of each metabolites. COT = cotinine; NNO = nicotine-1'-oxide; NCOT = norcortinine; NNIC = nornicotine; 3OHCOT = 3'-hydroxycotinine

Methods

EAE induction: Mice were immunized with the MOG_{35-55}/complete Freund's Adjuvant emulsion pertussis toxin in accordance with the manufacturer's instructions (EK-2110; Hooke Laboratories Inc., MA, USA). Body weights and clinical scores were assessed daily. Clinical scores between 0 to 5 were assigned, with a score of 5 indicating complete paralysis.

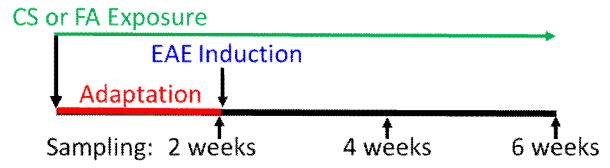
CS or FA Exposure
EAE Induction
Adaptation
Sampling: 2 weeks
4 weeks
6 weeks

Figure 2. Experimental timeline
Clinical score was monitored daily after EAE induction. Histological samples were collected at 2, 4, and 6 weeks. n = 8 per treatment group.

Histological analysis: Cervical and lumbar spinal cord samples were collected at 2, 4, and 6 weeks (Figure 2). Spinal cord samples were immersion-fixed in 4% paraformaldehyde and processed for DAPI and FluoroMyelin staining by NeuroScios/PsychoGenics Inc. (Austria). The number of lesions was quantified in a semi-automated manner by using Image Pro Premier (V 9.3).

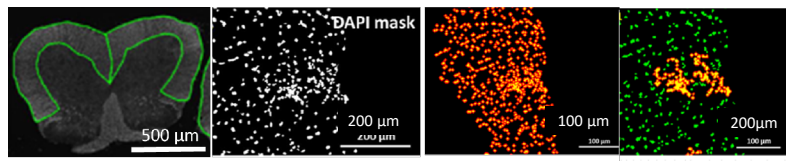
DAPI mask
500 µm
200 µm
100 µm
200µm

Figure 3: Histological quantification method
Dorsal white matter was segmented, and DAPI stained cells were identified. The cell borders were enlarged to form clusters.

Results

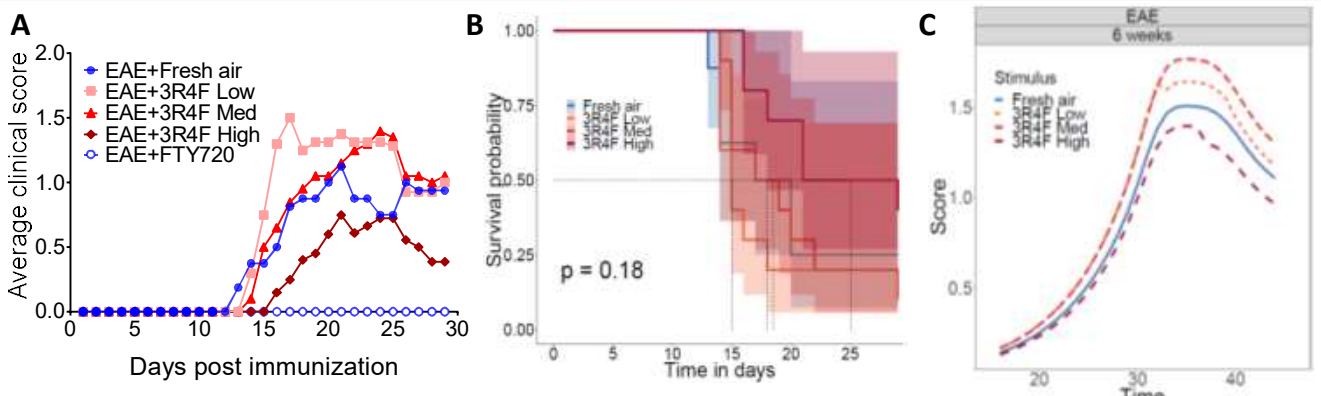
A
Average clinical score
EAE+Fresh air
EAE+3R4F Low
EAE+3R4F Med
EAE+3R4F High
EAE+FTY720
Days post immunization
B
Survival probability
p = 0.18
Time in days
C
EAE
6 weeks
Stimulus
Fresh air
3R4F Low
3R4F Med
3R4F High
Score
Time

Figure 4. Clinical scores for 3R4F-exposed EAE groups.
A) The average clinical score is shown for all EAE groups. FTY720 is shown as a positive control to block EAE. B) Clinical onset was analyzed by using non-parametric Kaplan–Meier estimator and log-rank tests. Exposure to high 3R4F CS significantly delayed the clinical onset of disease (p < 0.05), while low and medium 3R4F CS exposure had no significant effect in EAE-induced animals. C) Disease progression and severity were analyzed by using the Generalized Additive Model (GAM). Exposure to low and medium 3R4F CS significantly increased the clinical score (p < 0.05), while exposure to high 3R4F CS had no effect relative to the EAE group exposed to fresh air. There were no differences in disease progression among any of the groups, as indicated by the non-crossing, parallel slopes.

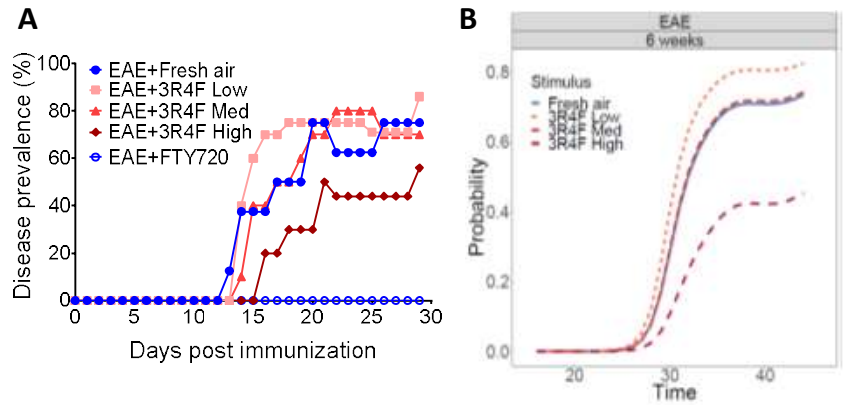
A
Disease prevalence (%)
EAE+Fresh air
EAE+3R4F Low
EAE+3R4F Med
EAE+3R4F High
EAE+FTY720
Days post immunization
B
EAE
6 weeks
Stimulus
Fresh air
3R4F Low
3R4F Med
3R4F High
Probability
Time

Figure 5. Disease prevalence for 3R4F-exposed EAE groups.
A) Descriptive data indicating the average daily probability of disease occurrence. B) GAM analysis of the data. Disease prevalence is the lowest for the high 3R4F exposure EAE group (p < 0.05). No significant change was observed in the low and medium 3R4F-exposed EAE groups.

Results

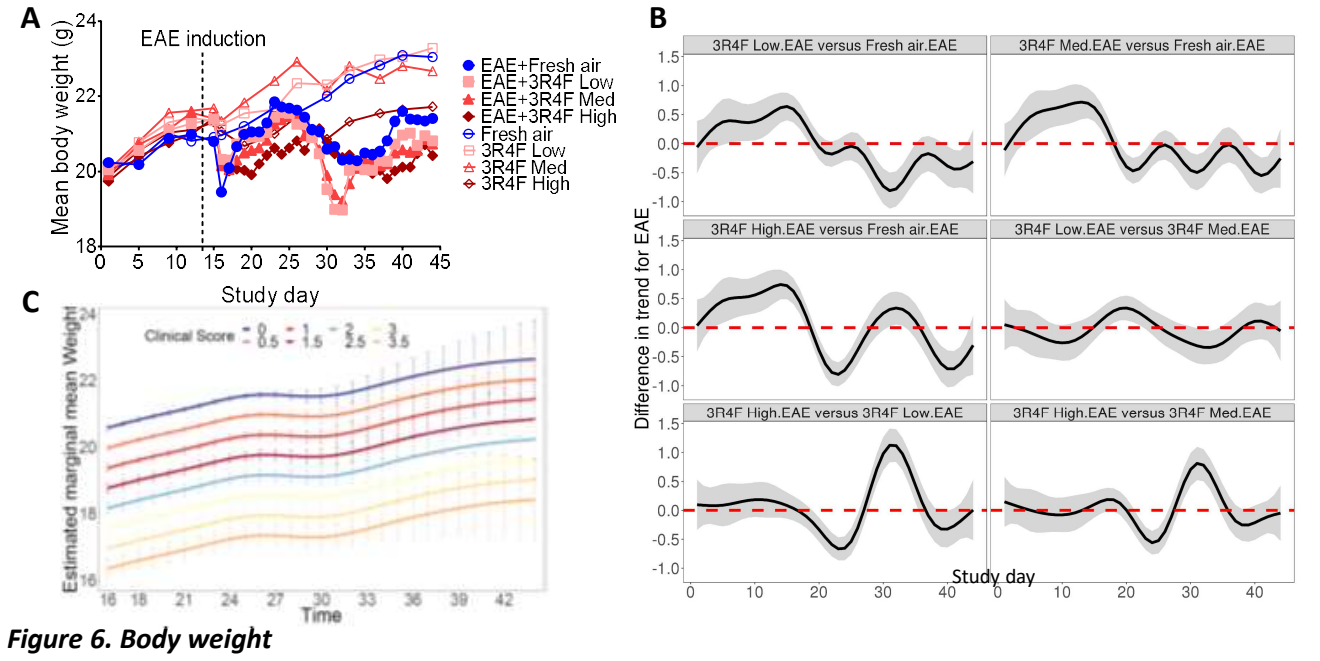
A
EAE induction
Mean body weight (g)
Study day
EAE+Fresh air
EAE+3R4F Low
EAE+3R4F Med
EAE+3R4F High
Fresh air
3R4F Low
3R4F Med
3R4F High
B
3R4F Low.EAE versus Fresh air.EAE
3R4F Med.EAE versus Fresh air.EAE
3R4F High.EAE versus Fresh air.EAE
3R4F Low.EAE versus 3R4F Med.EAE
3R4F High.EAE versus 3R4F Low.EAE
3R4F High.EAE versus 3R4F Med.EAE
Difference in trend for EAE
Study day
C
Estimated marginal mean Weight
Time

Figure 6. Body weight
A) Body weight in EAE-induced animals is reduced relative to non-EAE animals. B) Body weight fluctuates over the course of the study. The body weights of low and med 3R4F-exposure groups are similar. There are some differences in body weight progression depending on the dose of 3R4F. C) Body weight correlates well with the clinical score (p < 0.05), according to the results of GAM analysis.

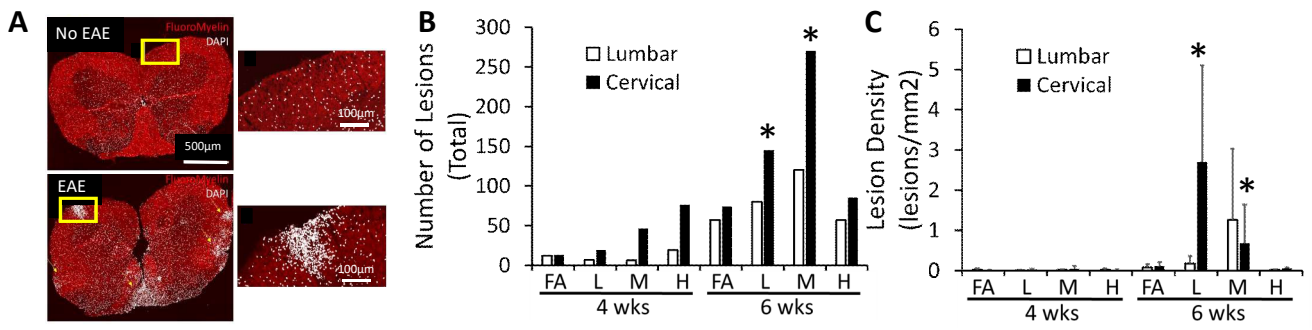
A
No EAE
EAE
500µm
100µm
B
Number of Lesions (Total)
Lumbar
Cervical
FA L M H FA L M H
4 wks
6 wks
C
Lesion Density (lesions/mm2)
Lumbar
Cervical
FA L M H FA L M H
4 wks
6 wks

Figure 7. Spinal cord pathology for the 3R4F-exposed EAE groups
A) Representative images of spinal cord sections labeled with DAPI (white) and FluoroMyelin (red) from non-EAE and EAE mice. B) The total number of lesions is significantly increased in the low (L) and medium (M), but not in the high(H) 3R4F-exposed EAE groups at 6 weeks (6 wks). C) Lesion density showing the same dose-dependent effect of 3R4F. The total number of lesions and clinical score were significantly correlated (p < 0.001), according to the results of Spearman's rank correlation analysis.*

Conclusions

- Exposure to low and medium CS doses exacerbated the clinical symptoms in EAE-induced mice, while it did not affect the probability of deterioration and symptom onset.
- Exposure to a high dose of CS had no significant effect on disease onset, severity, or progression, but it reduced the disease prevalence in EAE-induced mice.
- Spinal cord pathology was not evident at the onset of clinical symptoms.
- The spinal cord pathology before EAE-induction, at the onset of EAE symptoms, and at the end of the study correlated well with the clinical findings.
- Our results show that CS can worsen EAE symptoms and pathology in mice exposed to specific dose of CS before and after induction of EAE, which partially confirmed the findings in humans.
- The reasons for the observed dose-dependent effect of CS is unclear and requires further investigation.

References
1. O'Gorman, C., and Broadley, S.A. (2014). Journal of neurology 261, 1677-1683.
2. Wingerchuk, D.M. (2012). Therapeutic advances in neurological disorders 5, 13-22.

PMI SCIENCE
PHILIP MORRIS INTERNATIONAL

Society for Neuroscience, Chicago
October 19–23, 2019

Competing Financial Interest: The research described in this poster was sponsored by Philip Morris International. All authors are PMI employees.