Human organotypic nasal epithelial tissue culture as an *in vitro* model to evaluate effects of cigarette smoke of a candidate Modified Risk Tobacco Product: the Tobacco Heating System 2.2

C. Mathis, M. Cabanski, S. Frentzel, D. Kuehn, S. Majeed, C. Merg, A. Elamin, E. Guedj, R. Dulize, D. Peric, K. Trivedi, A. Benyagoub, Y. Xiang, F. Martin, P. Leroy, N.V. Ivanov, M.C. Peitsch, J. Hoeng

Philip Morris International, Philip Morris Products S.A., Neuchâtel, Switzerland

INTRODUCTION

Exposure to cigarette smoke (CS) is a major risk of developing serious diseases such as lung cancer, chronic obstructive pulmonary disease or cardiovascular disease.

The development of new tobacco products that could reduce such health impact is ongoing and requires careful safety assessment strategy.

To investigate the effect of the aerosol generated by Philip Morris International's candidate Modified Risk Tobacco Product (MRTP), named Tobacco Heating System 2.2 (THS2.2), an *in vitro* model mimicking the human nasal epithelium was exposed for 28 min at the air liquid interface to various doses. In parallel, exposure with air (sham control) or with mainstream smoke from 3R4F cigarette (at doses where nicotine level within the aerosol are matching those of THS2.2 product) was performed. Various endpoints (cytotoxicity, CYP1A1/1B1 enzyme activity, inflammatory markers release, morphological and gene expression changes) were collected at different time after exposure (4, 24, 48 and 72h) to identify and compare the dose- and time-dependent effect of each exposure conditions.

By using systems toxicology-based risk assessment approaches combining computable biological network models and gene expression changes (1), we compared the molecular perturbations in both 3R4F cigarette and MRTP exposure conditions.

While significant effect was quantified over different post-exposure time points in the networks representing cell death, inflammation, proliferation and cellular stress after cigarette smoke (CS) exposure, the impact of THS2.2 exposure (at similar dose) was closer to sham controls and mostly limited at the earliest time point (4h). The results of all the additional endpoints (see figure 4: histological assessment and figure 7: inflammatory markers release, CYP1A1/1B1 activity and cytotoxicity) measured during this study support a reduced impact of THS2.2 acute exposure on the nasal epithelial tissue culture compared to 3R4F cigarette exposure.

MATERIALS and METHODS

Figure 1: Schematic representation of the VITROCELL® 24/48 exposure system: A climatic chamber contains an exposure module (red frame) where up to 48 wells can be exposed simultaneously to up to 8 dilutions of an aerosol/smoke.

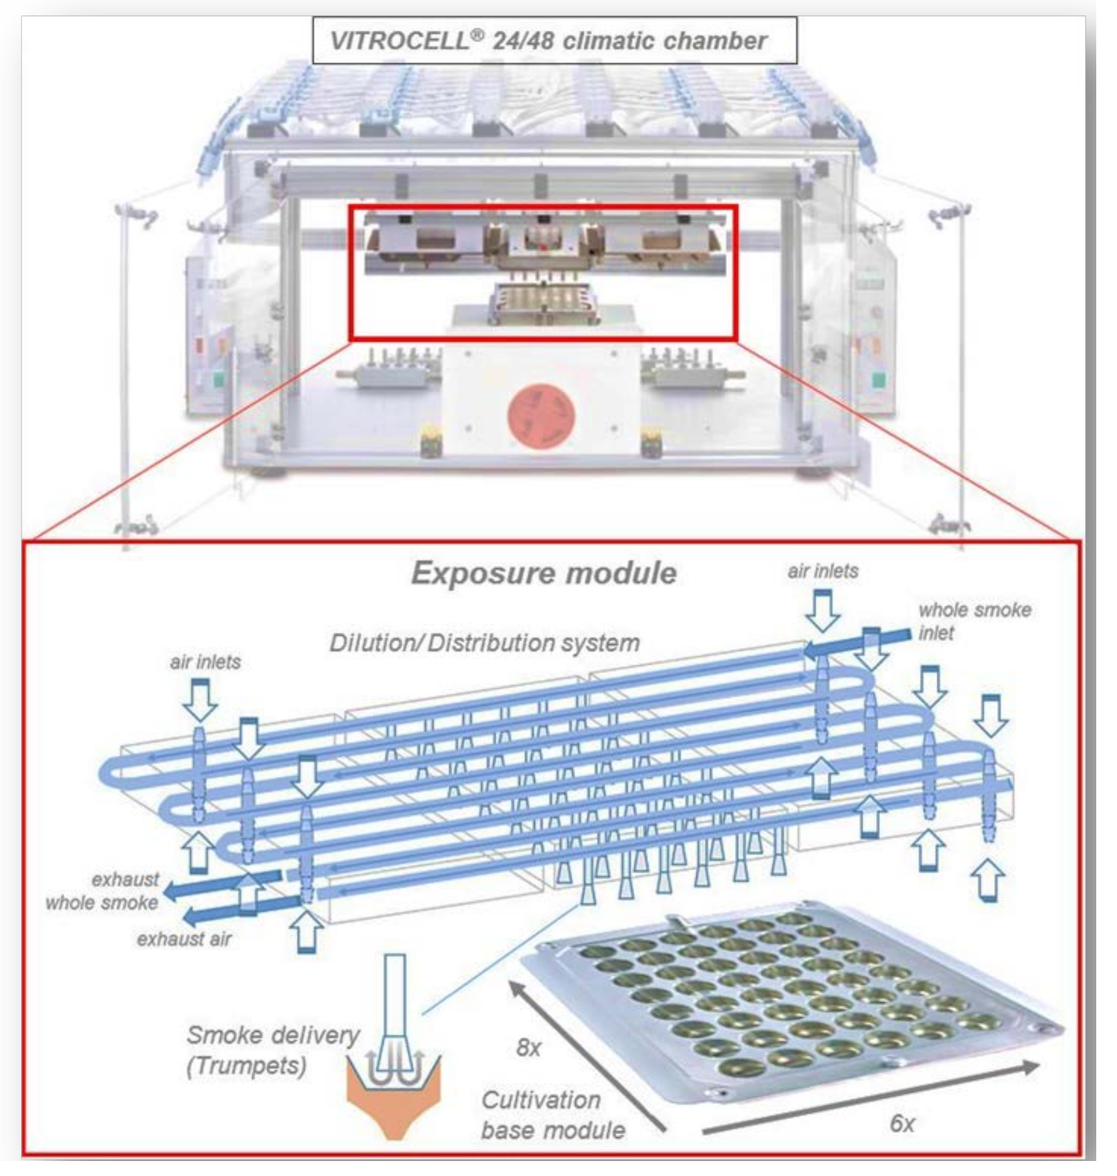

VITROCELL® 24/48 climatic chamber
Exposure module
Dilution/Distribution system
air inlets
whole smoke inlet
exhaust whole smoke
exhaust air
Smoke delivery (Trumpets)
Cultivation base module
8x
6x

Figure 2: (A) Experimental design. Human nasal Mucilair™ tissues (Epithelix – donor: 30 years-old male non smoker) were directly exposed at the air liquid interface for 28 min to diluted CS from 3R4F (reference cigarettes obtained from the University of Kentucky) or to 60% humidified air (air-exposed controls) or to diluted THS2.2 aerosol. After exposure, tissue inserts were incubated with fresh culture medium for 4h, 24h, 48h or 72h before measuring various endpoints. TEER: Trans Epithelial Electrical Resistance. AK: Adenylate kinase. **(B) Nicotine levels** were measured in the aerosol at different dilutions (%: vol/vol with air) of 3R4F smoke and THS2.2 aerosol using the extrelute method. Bars are means ± SEM (n=3-15).

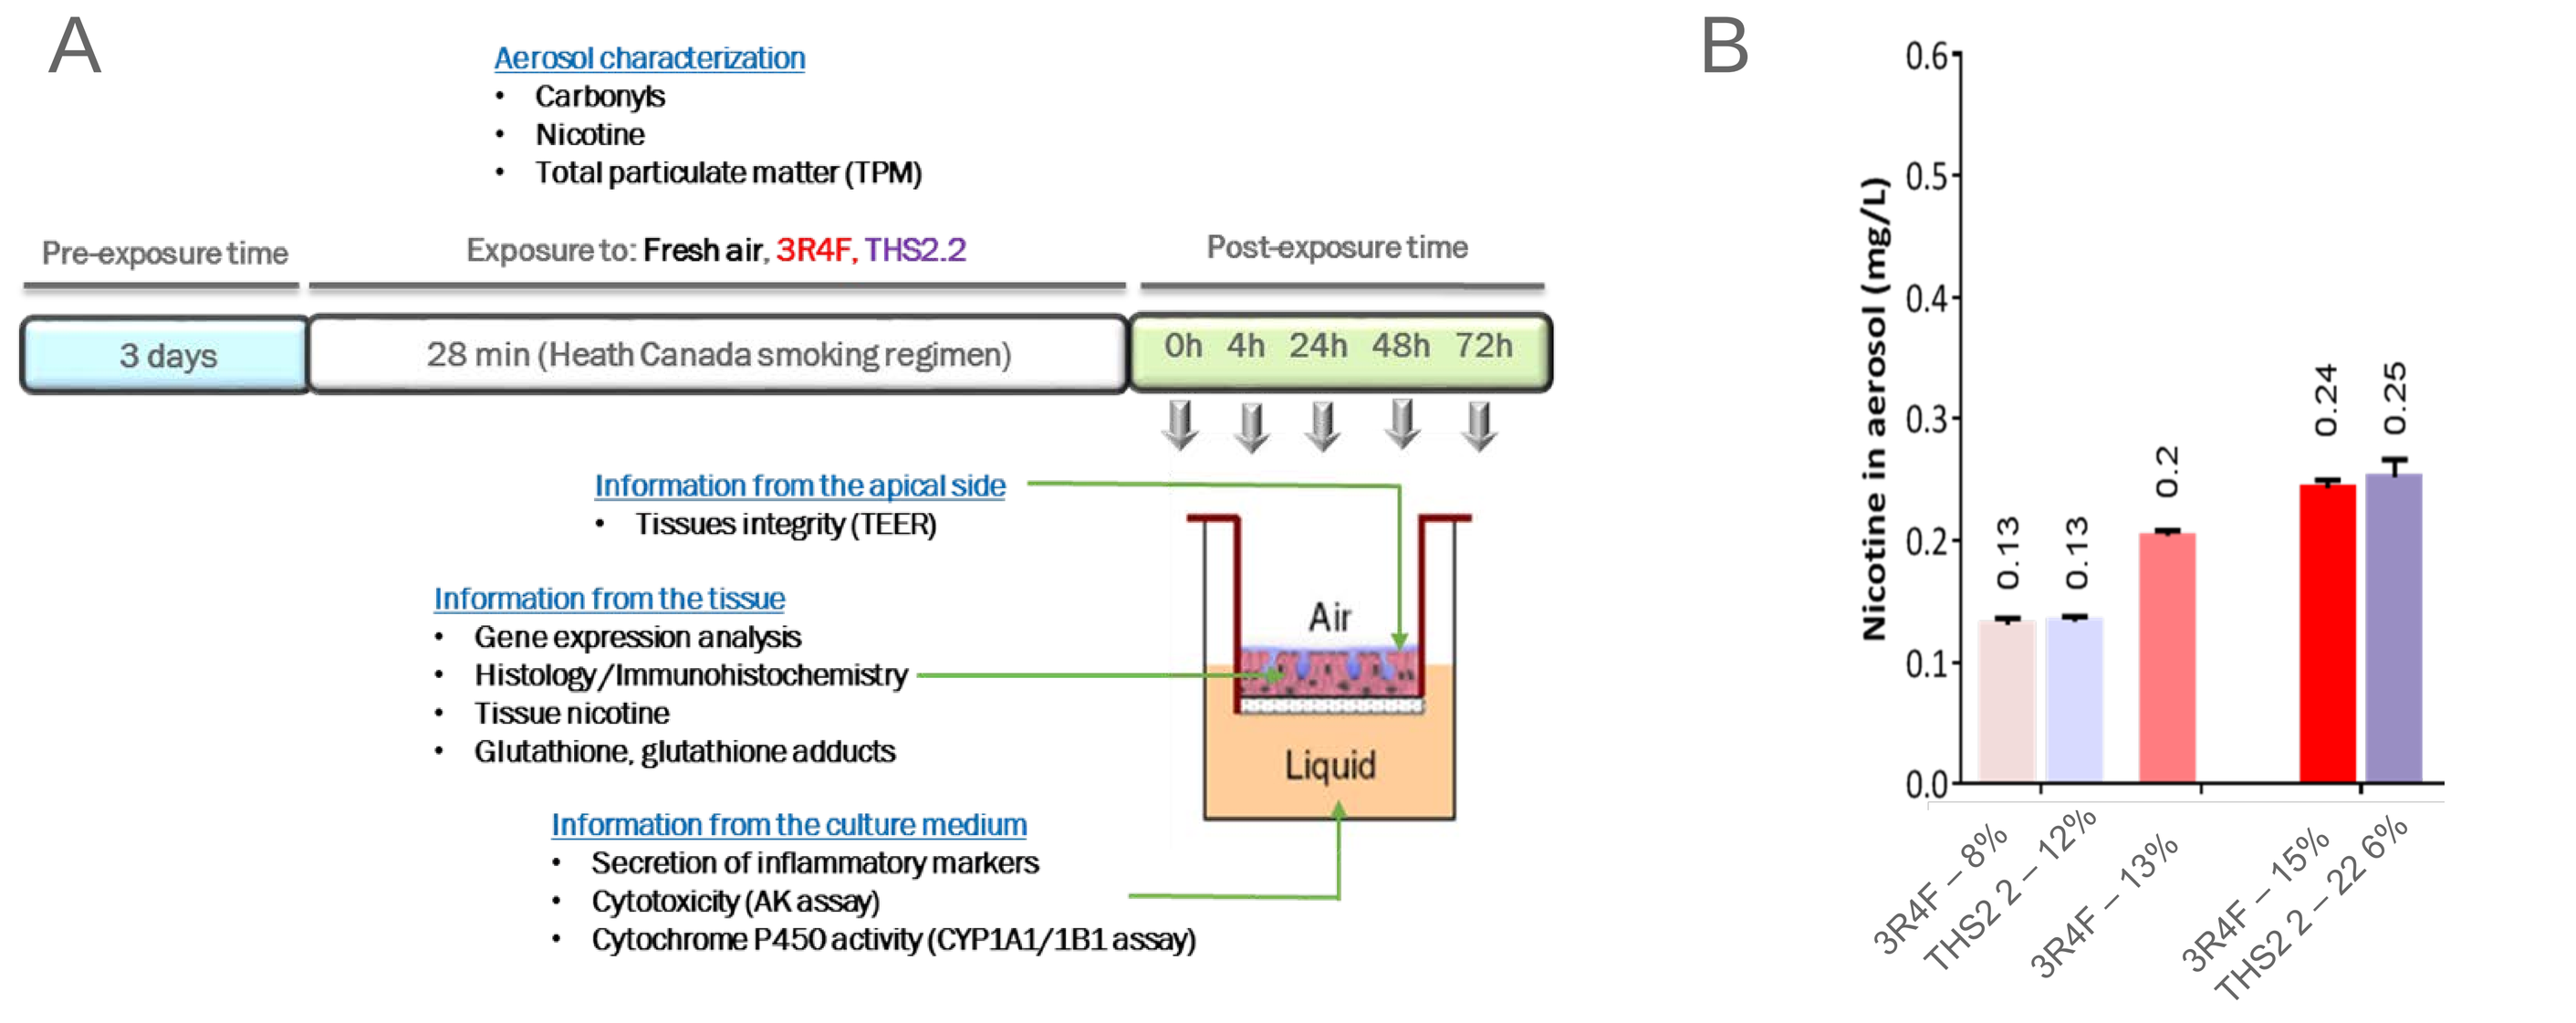

A
Aerosol characterization
• Carbonyls
• Nicotine
• Total particulate matter (TPM)
Pre-exposure time
Exposure to: Fresh air, 3R4F, THS2.2
Post-exposure time
3 days
28 min (Heath Canada smoking regimen)
0h 4h 24h 48h 72h
Information from the apical side
• Tissues integrity (TEER)
Information from the tissue
• Gene expression analysis
• Histology/Immunohistochemistry
• Tissue nicotine
• Glutathione, glutathione adducts
Information from the culture medium
• Secretion of inflammatory markers
• Cytotoxicity (AK assay)
• Cytochrome P450 activity (CYP1A1/1B1 assay)
Air
Liquid
B
Nicotine in aerosol (mg/L)
0.13
0.13
0.2
0.24
0.25
3R4F – 8%
THS2.2 – 12%
3R4F – 13%
3R4F – 15%
THS2.2 – 22.6%

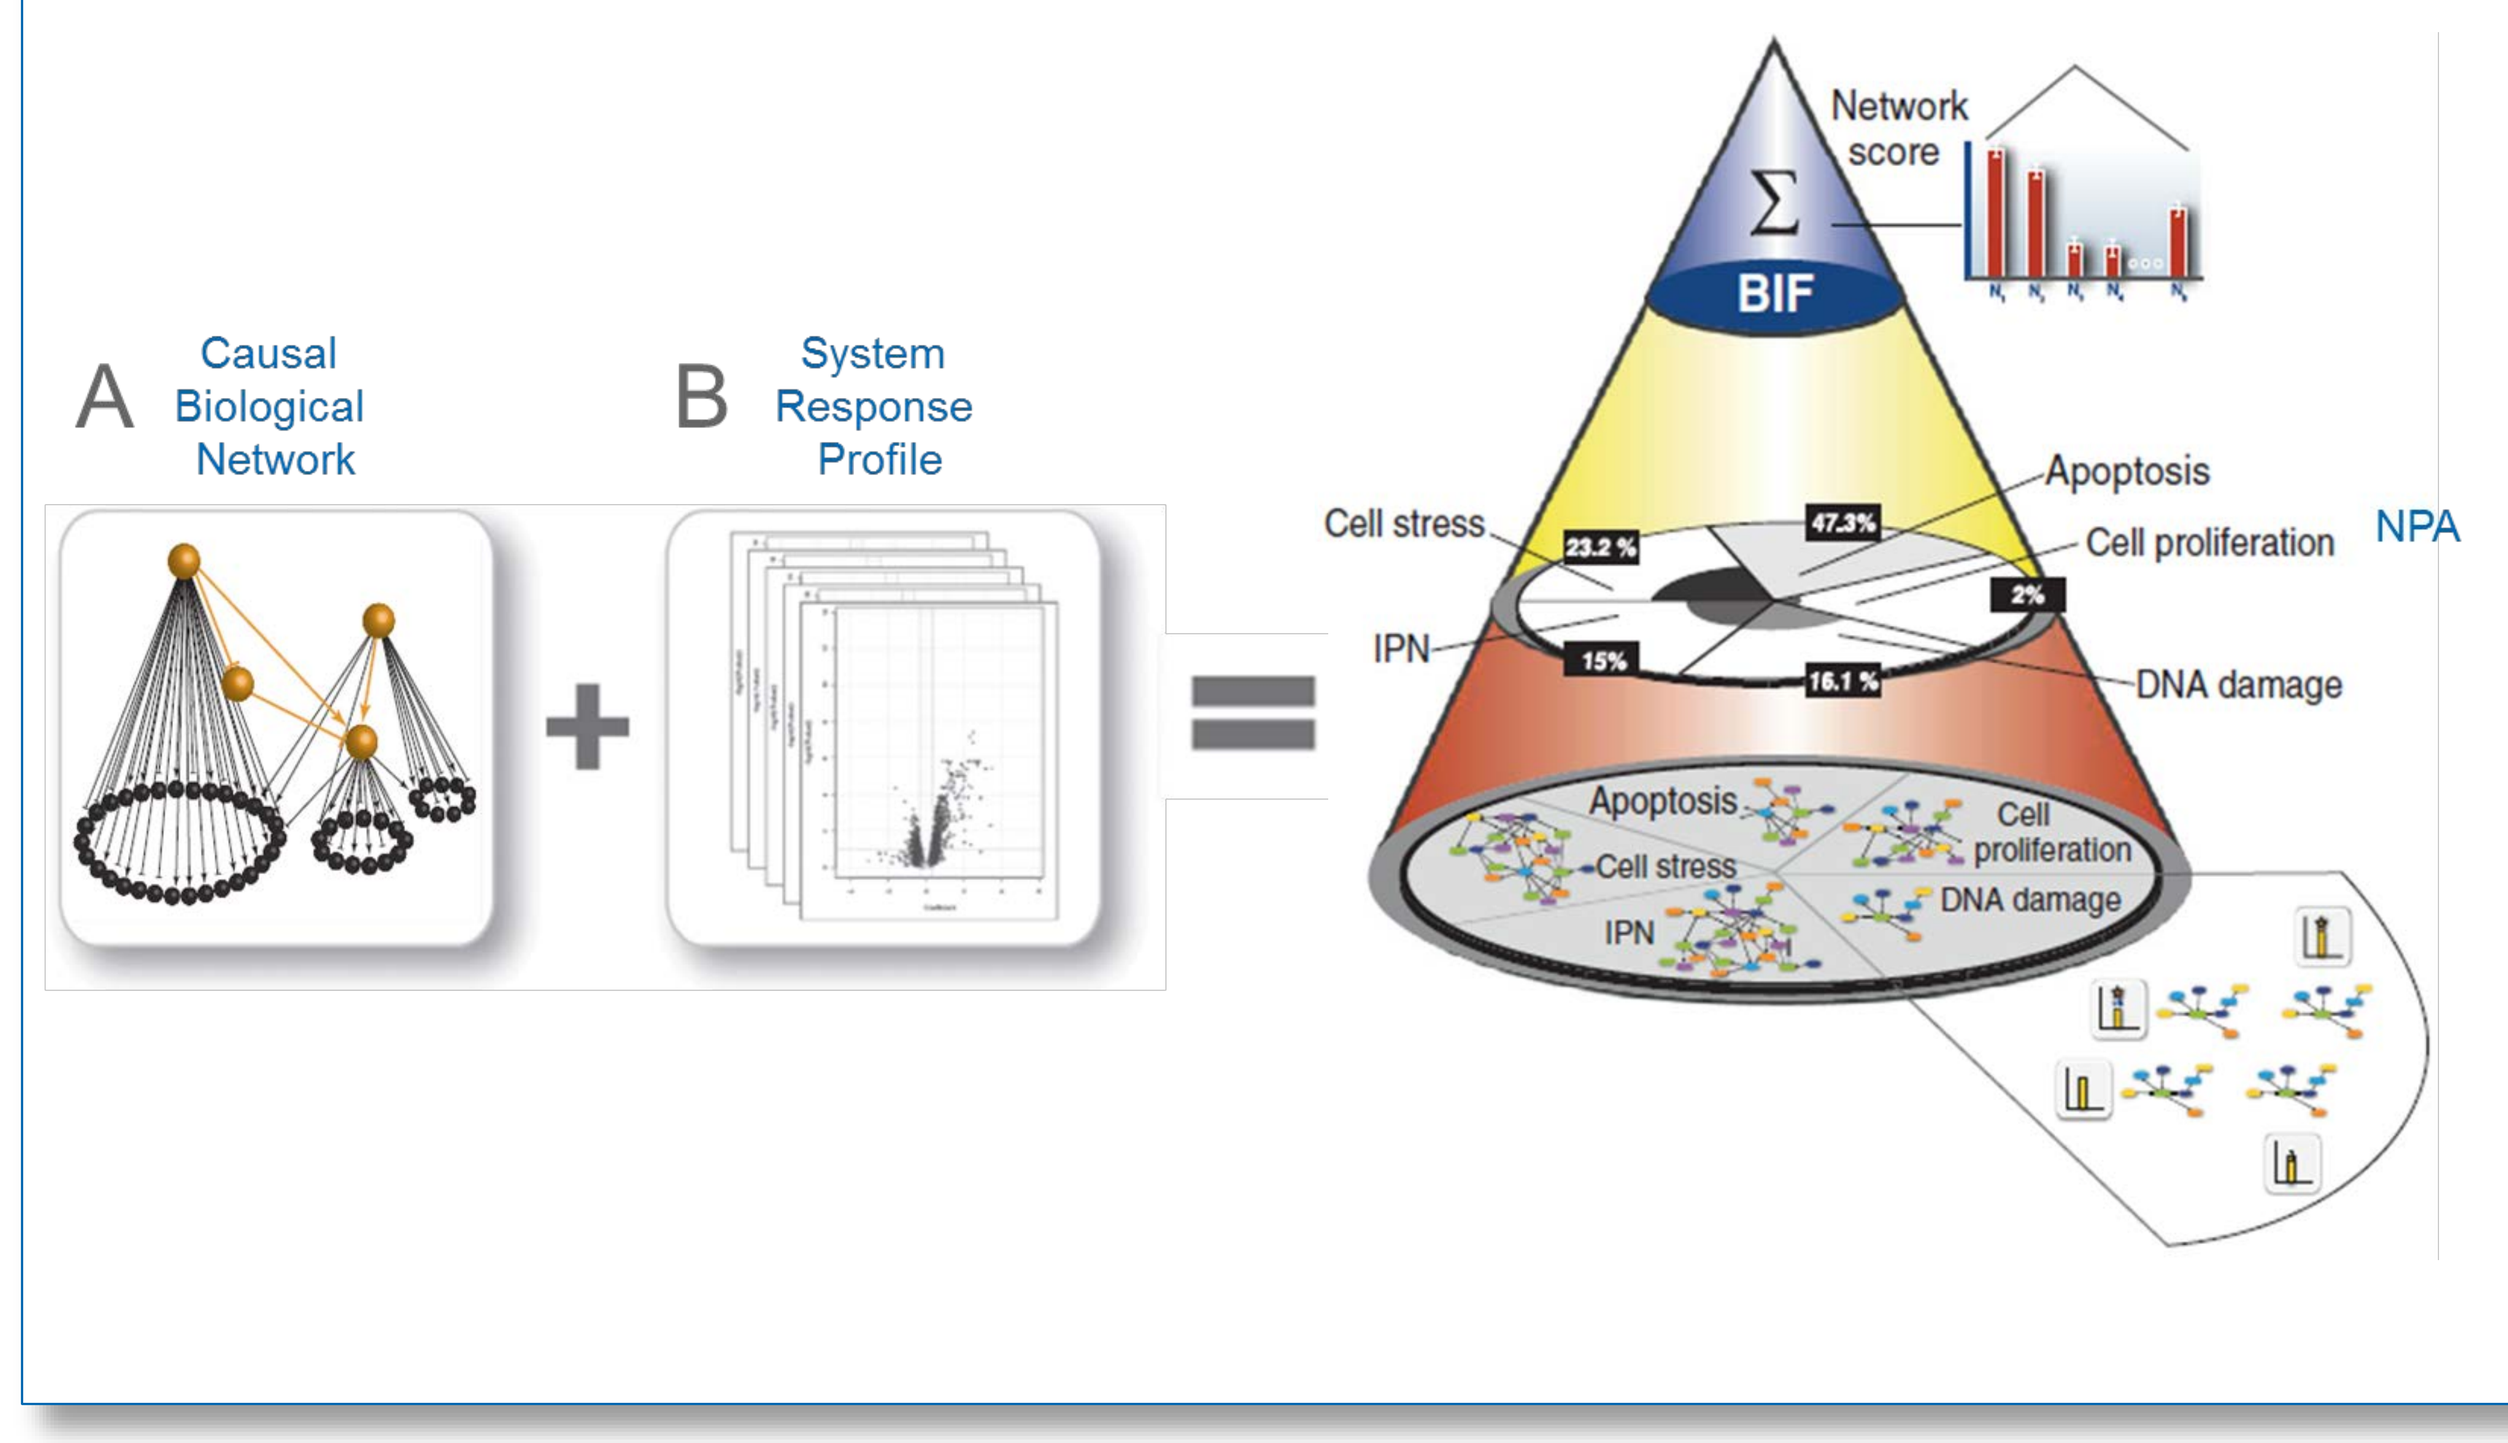

A Causal Biological Network
B System Response Profile
Network score
BIF
Cell stress
IPN
Apoptosis
Cell proliferation
DNA damage
NPA
47.3%
23.2 %
2%
15%
16.1 %

Figure 3: (A) The causal biological networks are describing biological processes or mechanisms (e.g., Cell Proliferation (2), Cell Stress (3), DNA damage and Apoptosis (4) or Inflammation (5)). They are composed of backbone nodes (big grey balls) connected by causal directional relationships (= edges) derived from an evidence line extracted from literature. Differential expression of genes (small black balls) are experimental evidences for the activation of upstream backbone node. (B) System response profiles (=differentially expressed genes) are translated into Network Perturbation Amplitude (NPA) scores (6) for each biological networks and sub-networks allowing a higher granularity of the biological interpretation of the dataset. The Biological Impact Factor (BIF) (7) is computed by aggregating NPA scores. It represents a hollistic score that describes the system-wide effect of all biological processes perturbed after exposure.

RESULTS

The morphology of THS2.2-exposed nasal Mucilair™ inserts is more comparable to air-exposed inserts then to 3R4F-exposed inserts at comparable nicotine dose

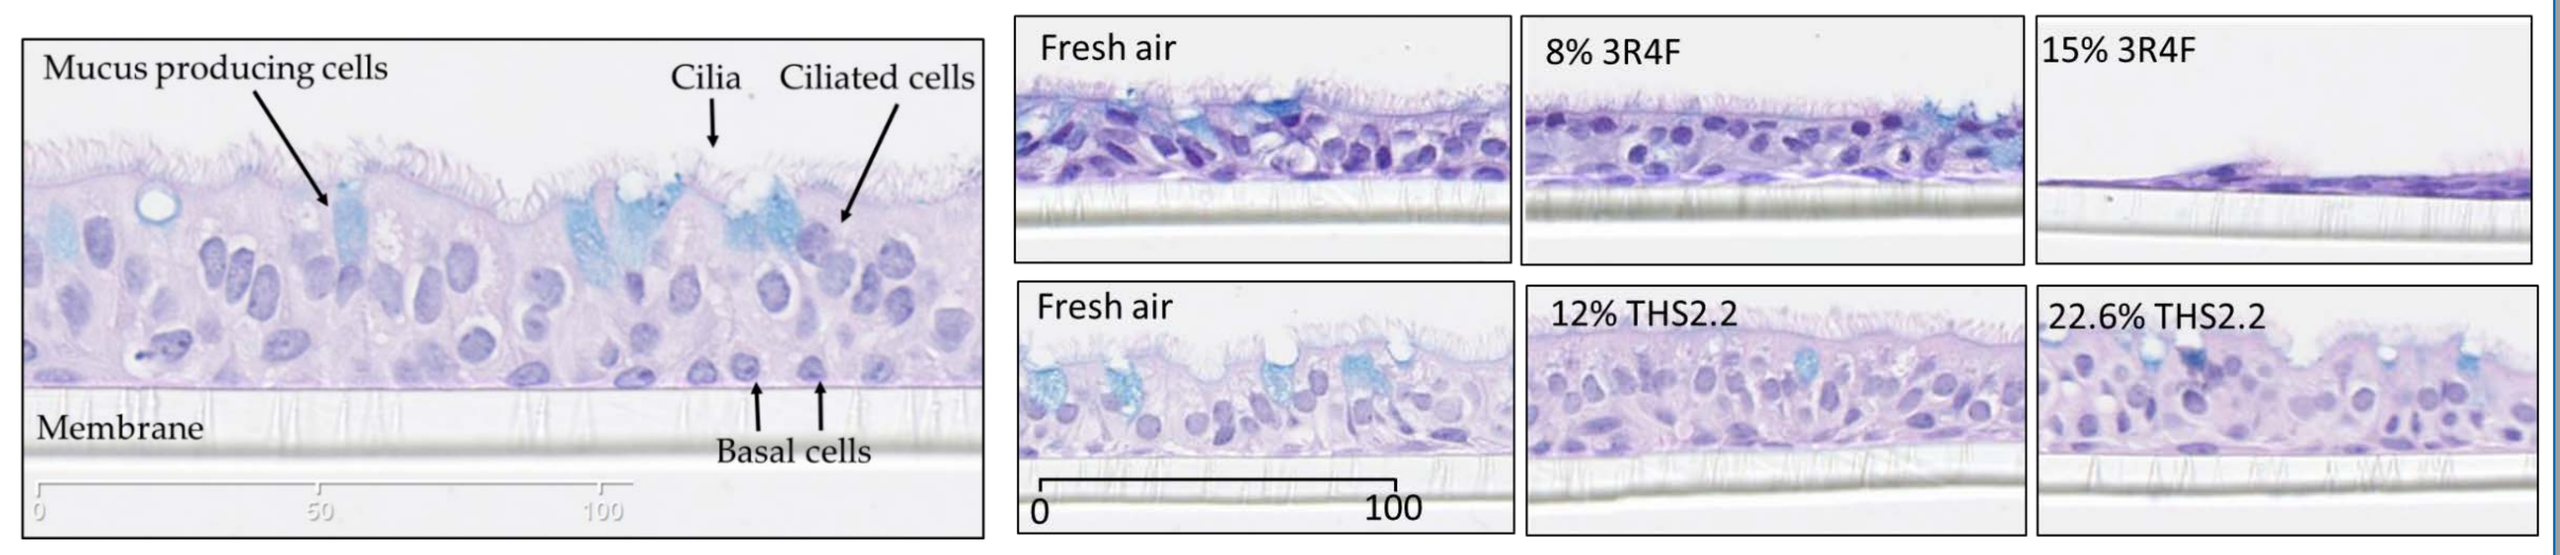

Mucus producing cells
Cilia
Ciliated cells
Membrane
Basal cells
Fresh air
8% 3R4F
15% 3R4F
Fresh air
12% THS2.2
22.6% THS2.2
0
100

Figure 4: Morphological assessment using Hematoxylin/Eosin/Alcian blue staining of paraffin-embedded sections. (A) Untreated nasal Mucilair™ tissue culture shows a pseudostratified epithelium composed of mucus producing cells, cilia, ciliated cells, and basal cells. (B) Representative pictures of stained sections from tissue inserts two days after a 28 min exposure to air, 8%/15% of 3R4F smoke or to corresponding doses (at the nicotine level) of THS2.2 aerosol (12%/22.6%).

- **A clear dose- and time-dependent effect is observed on gene expression in 3R4F-exposed nasal Mucilair™ inserts compared to air-exposed controls.**
- **THS2.2 exposure induces only an early dose-dependent response in exposed nasal Mucilair™ inserts.**
- **At comparable dose (nicotine level) and post-exposure time points, 3R4F exposure has always a stronger effect on gene expression than THS2.2 exposure.**

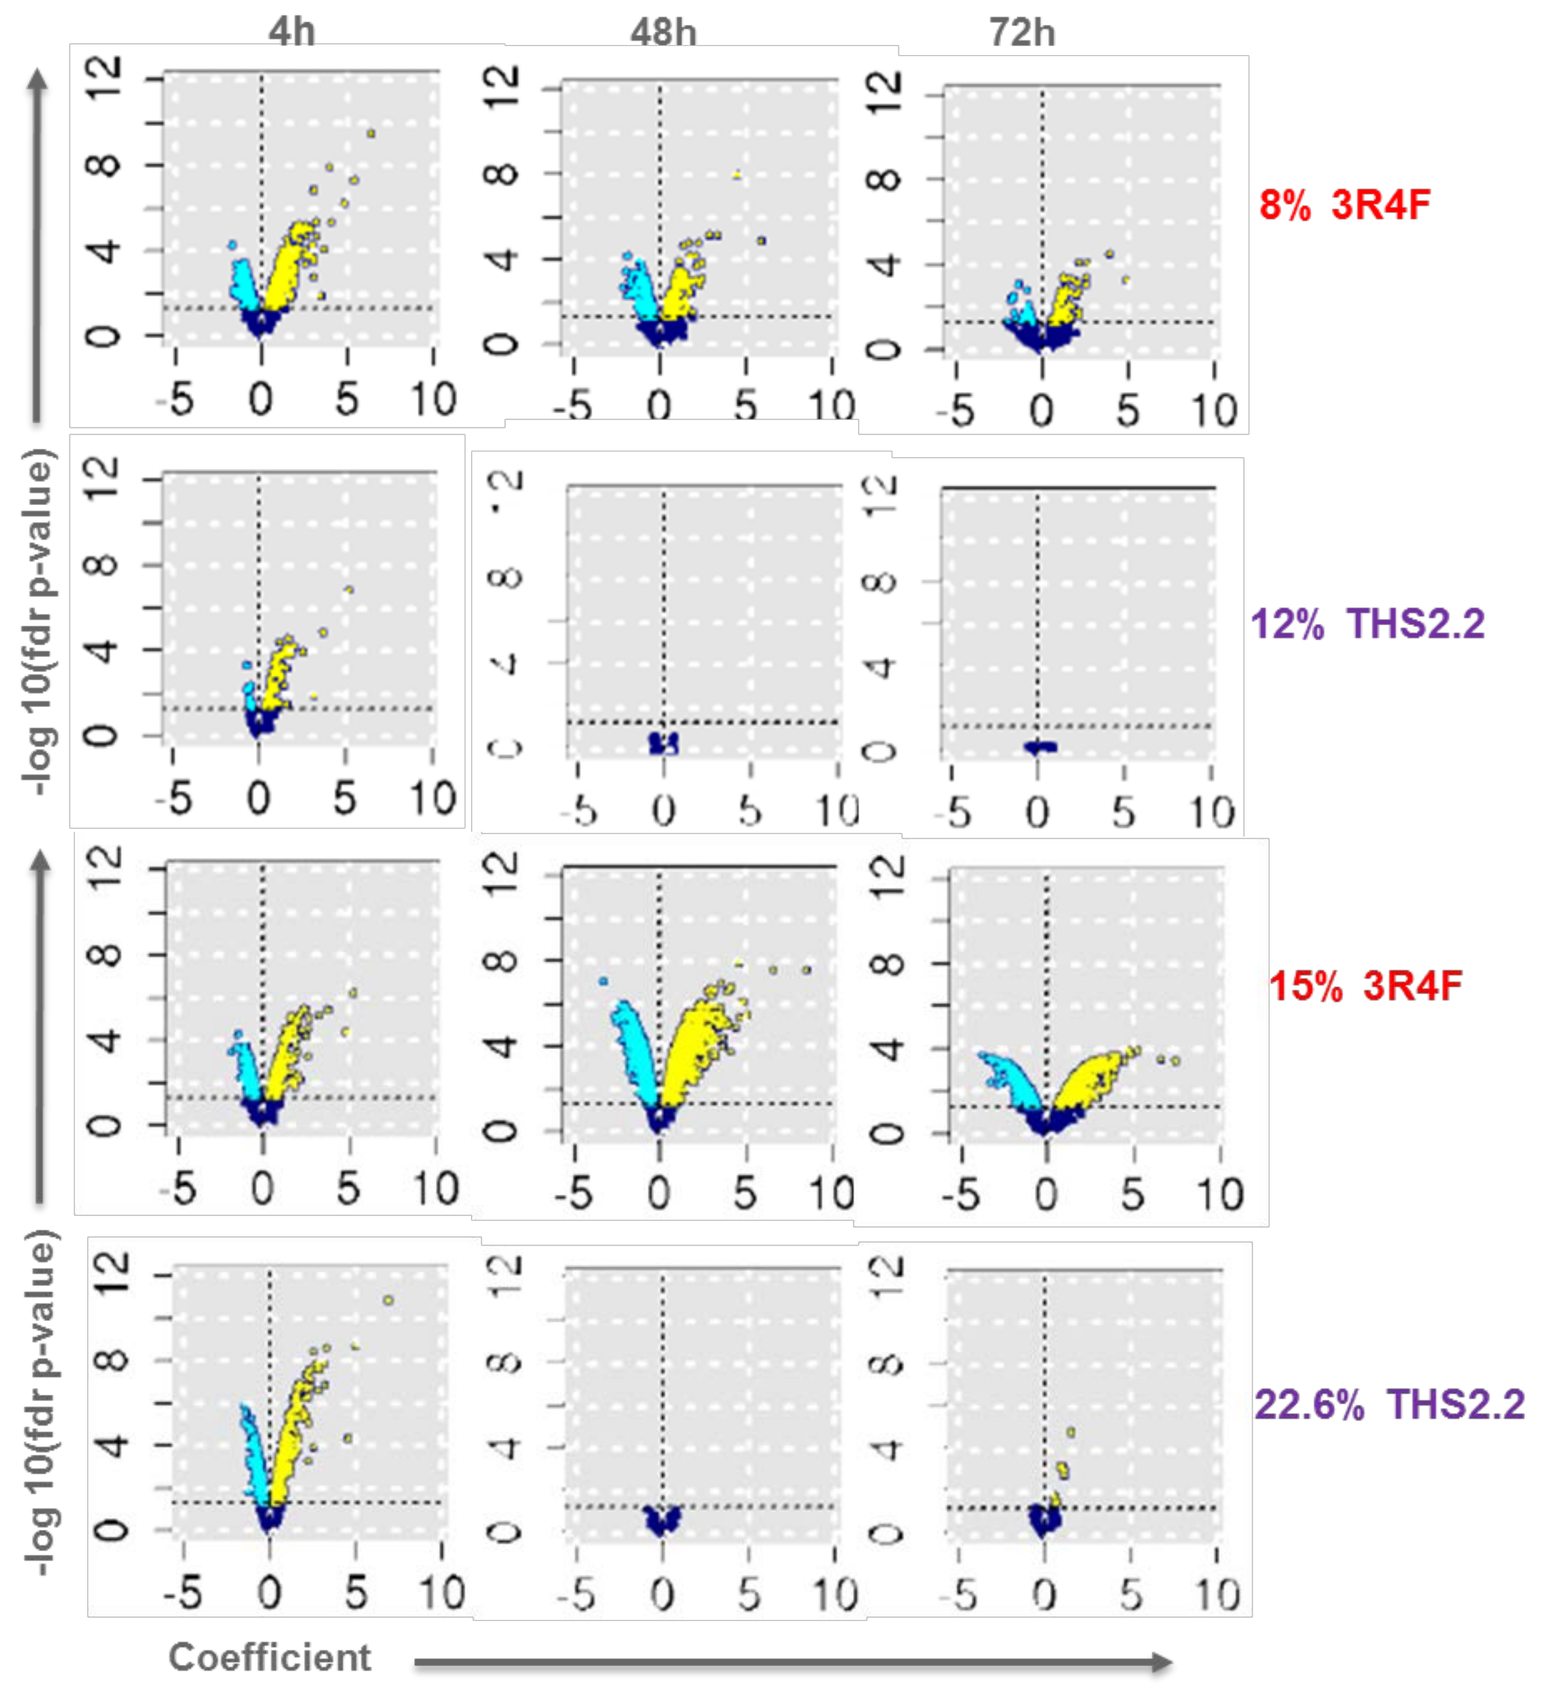

4h
48h
72h
8% 3R4F
12% THS2.2
15% 3R4F
22.6% THS2.2
-log 10(fdr p-value)
Coefficient

Figure 5: Dose- and time-dependent effect on gene expression of 3R4F- and THS2.2-exposed nasal Mucilair™ inserts. Volcano plots representing global differentially expressed genes (light blue dots: down-regulated; yellow dots: up-regulated; dark blue dots: below fdr p-value of 0.05) from MucilAir™ inserts 4h, 48h and 72h after exposure to comparable doses (at the nicotine level) of 3R4F smoke or THS2.2 aerosol versus air controls. Coefficient presents log2-based fold change. fdr: false discovery rate.

Reduced dose-dependent network perturbations of THS2.2-exposed nasal Mucilair™ compared to 3R4F-exposed tissue at comparable level of nicotine in the aerosol and at different post-exposure time points

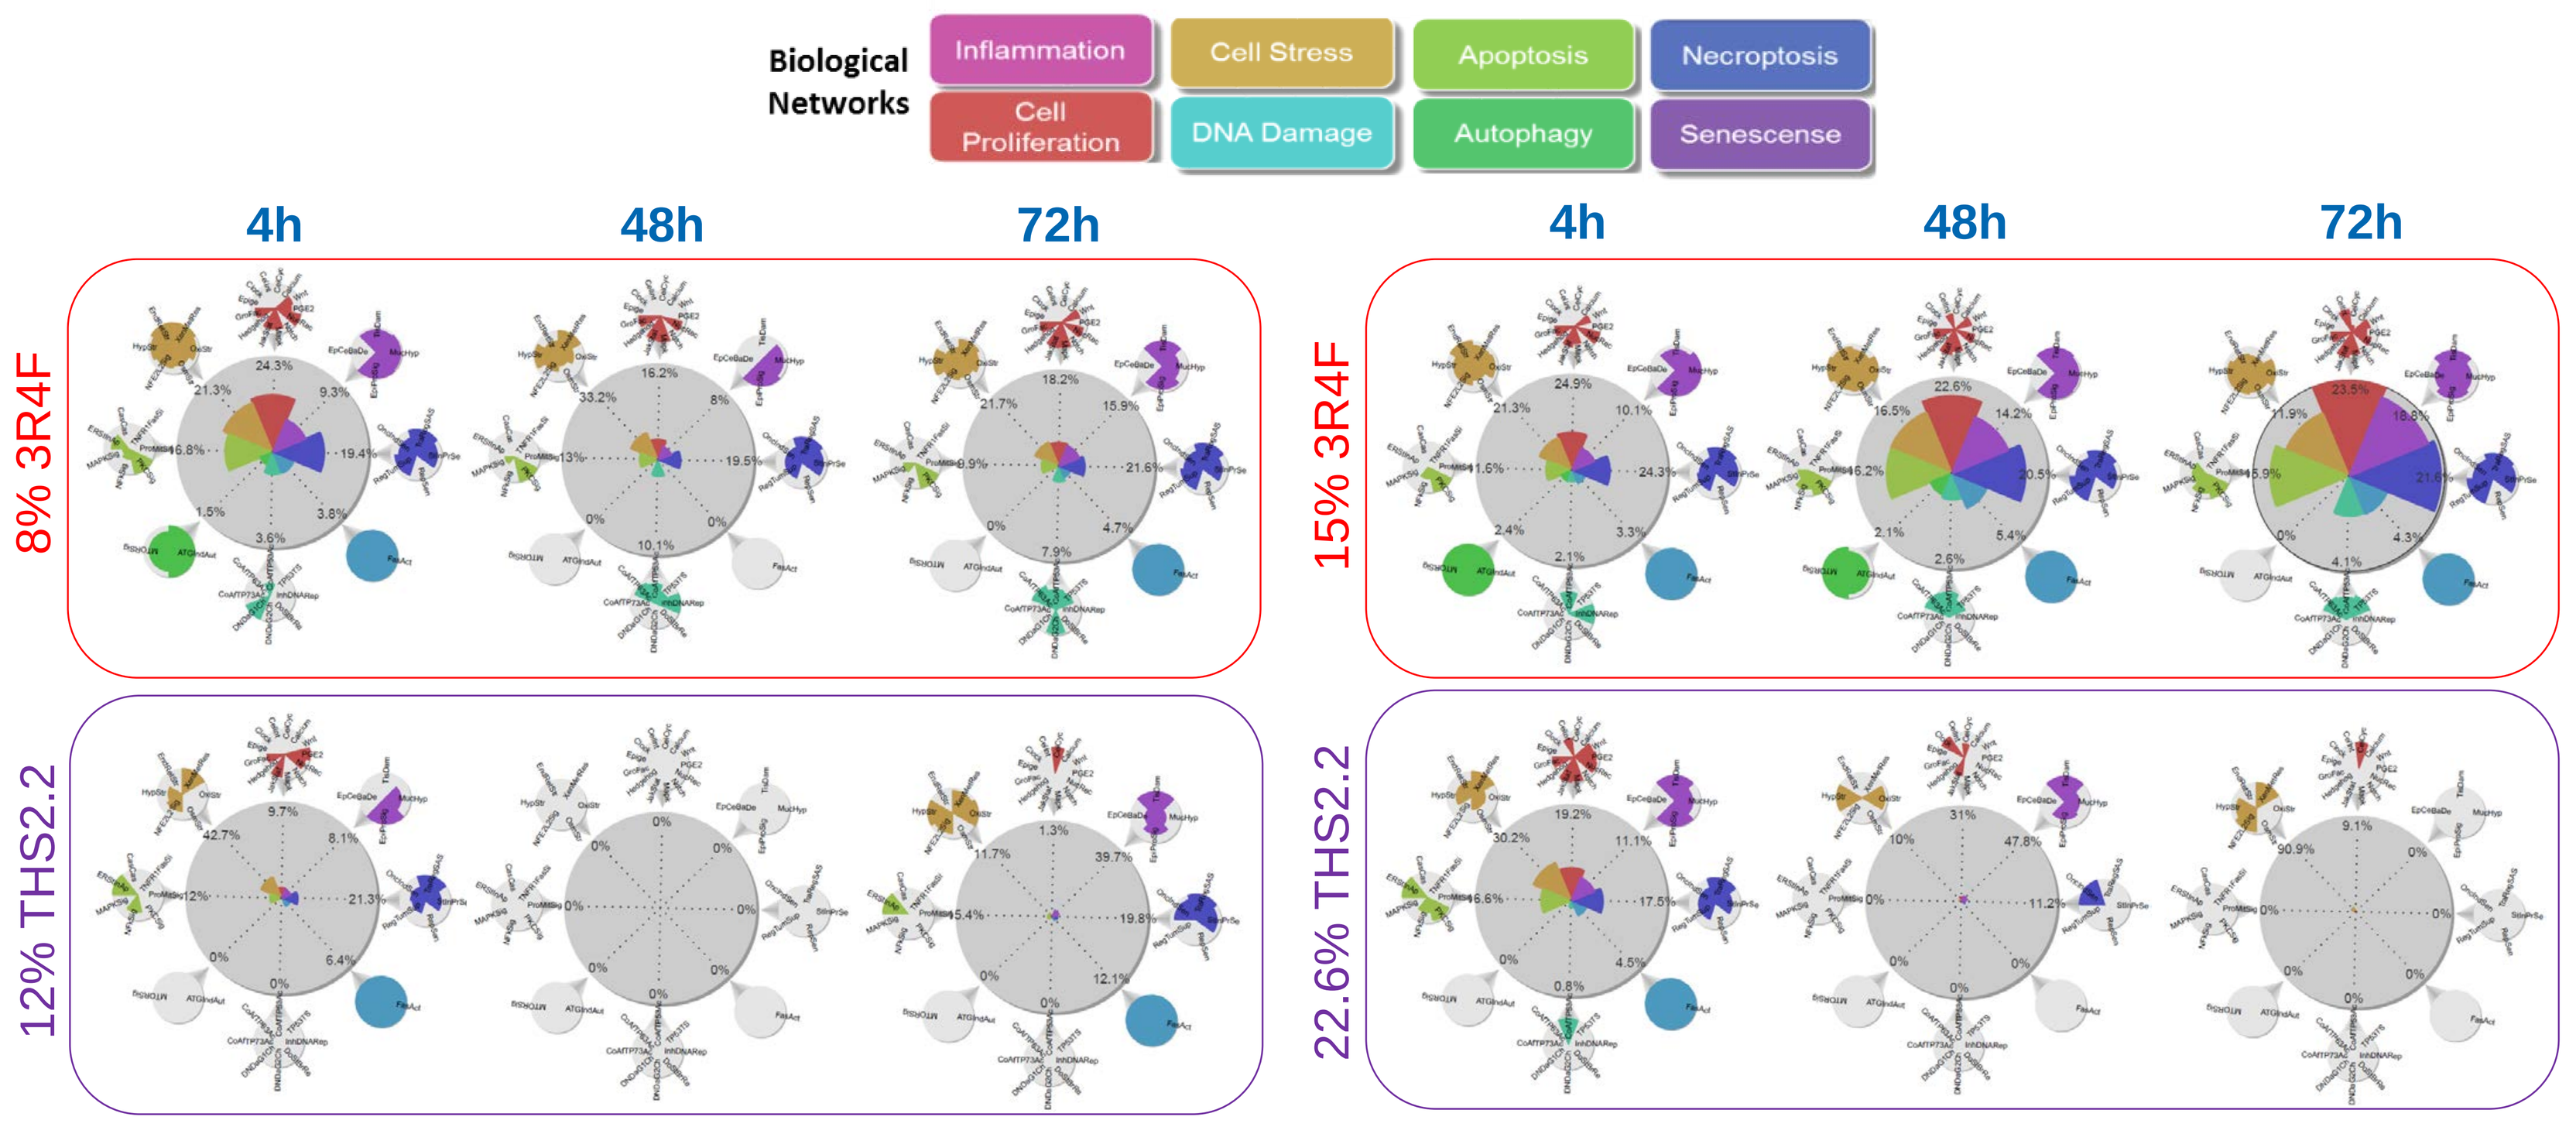

Biological Networks
Inflammation
Cell Stress
Apoptosis
Necroptosis
Cell Proliferation
DNA Damage
Autophagy
Senescense
4h
48h
72h
8% 3R4F
15% 3R4F
12% THS2.2
22.6% THS2.2

Figure 6: Quantification of the dose- and time-dependent perturbations of different biological networks (indicated in the rainbow colors boxes) using gene fold-changes as an input and the NPA method (6) as a computational approach. The surface area of each slide is proportional to the contribution within a particular treatment of each network perturbation (shown in percentage). It was further adjusted by the relative Biological Impact Factor (BIF) (7) for treatments compared with the reference, so the sum of the slice areas for each treatment equals the BIF (7) of the treatment. The pie chart represents the distribution of the sum of the contributions of each network across all treatment groups.

- **The meta-analysis shows a significant positive dose-response after 3R4F exposure in the secretion of TIMP1, MMP1, CCL20, CXCL8, EGF, sICAM1, IL1A, VEGFA and in the AK activity and a negative dose-response for CYP1A1/1B1 activity.**
- **The overall effect of THS2.2 exposure was significantly reduced compared to 3R4F and close to air-exposed inserts.**

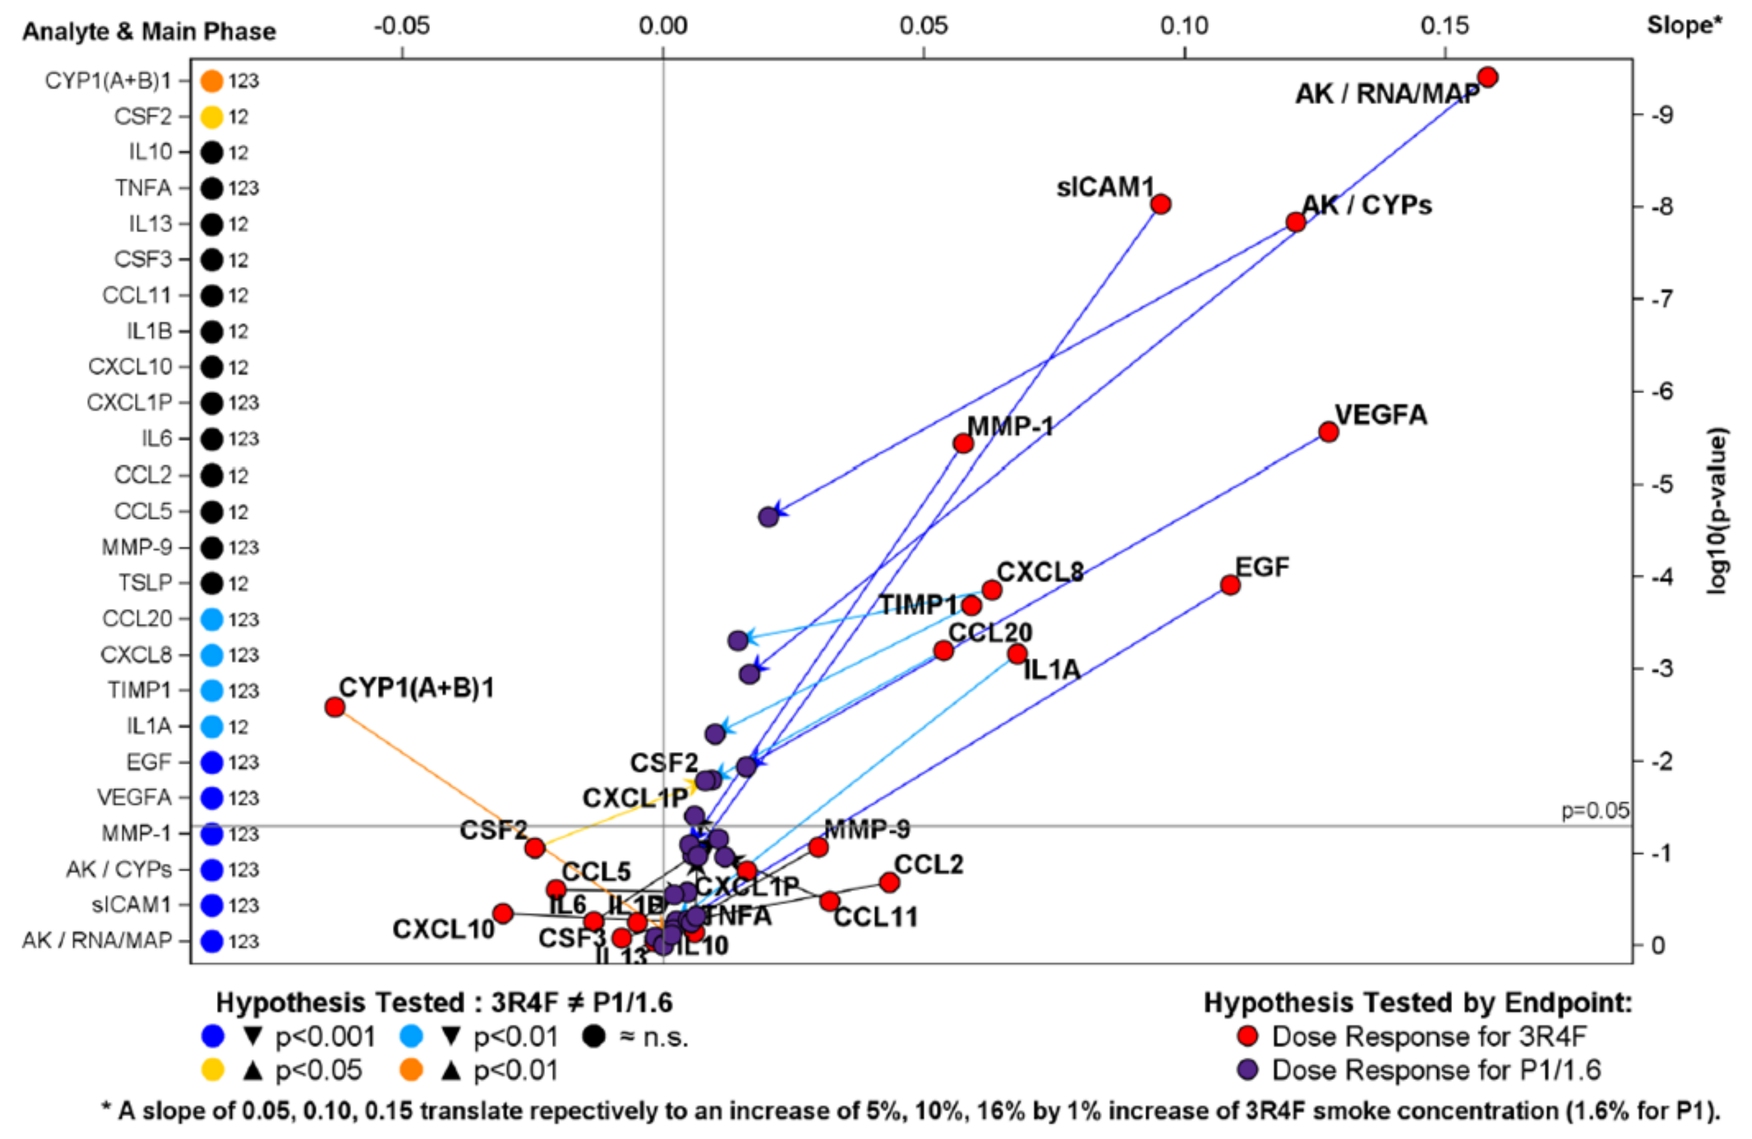

Analyte & Main Phase
Slope*
log10(p-value)
p=0.05
AK / RNA/MAP
sICAM1
AK / CYPs
MMP-1
VEGFA
CXCL8
TIMP1
CCL20
EGF
IL1A
CYP1(A+B)1
CSF2
CXCL1P
MMP-9
CCL2
CCL5
CCL11
CXCL10
Hypothesis Tested : 3R4F ≠ P1/1.6
Hypothesis Tested by Endpoint:
Dose Response for 3R4F
Dose Response for P1/1.6
* A slope of 0.05, 0.10, 0.15 translate repectively to an increase of 5%, 10%, 16% by 1% increase of 3R4F smoke concentration (1.6% for P1).

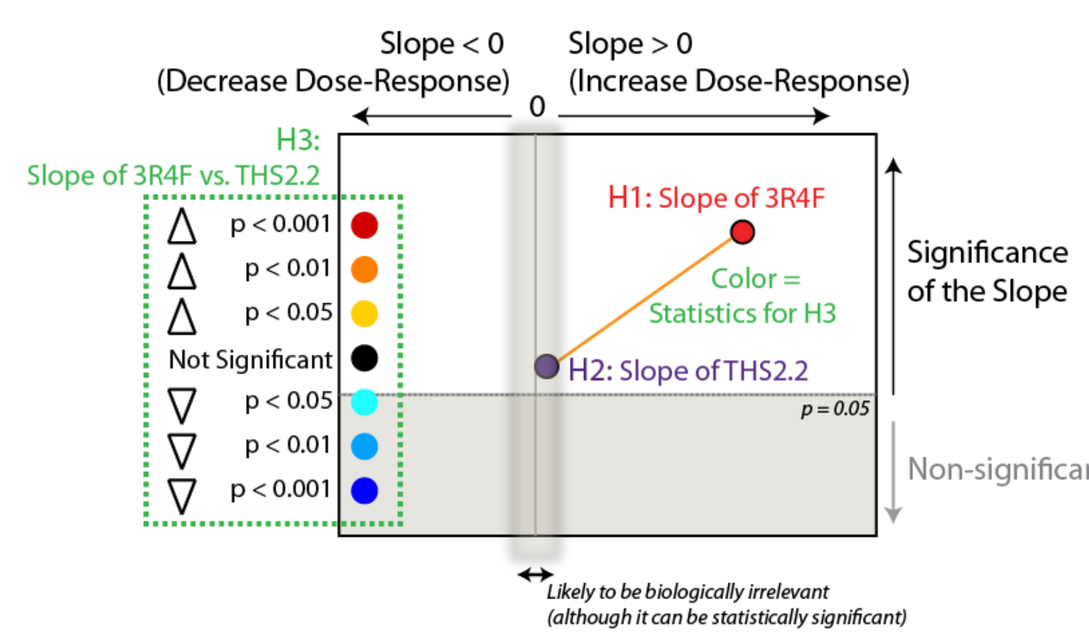

Slope < 0 (Decrease Dose-Response)
Slope > 0 (Increase Dose-Response)
H3: Slope of 3R4F vs. THS2.2
H1: Slope of 3R4F
H2: Slope of THS2.2
Color = Statistics for H3
Significance of the Slope
Non-significant Slope
p < 0.001
p < 0.01
p < 0.05
Not Significant
Likely to be biologically irrelevant (although it can be statistically significant)

Figure 7: Meta-analysis of the effects (measurement of secreted individual inflammatory markers, CYP1A1/1B1 activity assay, AK assay) observed 48h after exposure to all doses of 3R4F or THS2.2 versus air-exposed controls.

CONCLUSIONS

- Human nasal Mucilair™ tissue cultures were successfully exposed for 28min in parallel to air and to diluted smoke/aerosol from 3R4F cigarette and THS2.2 at similar nicotine concentration (see figure 2B).
- A dose- and a time-dependent effect was observed after 3R4F exposure at gene expression level (see figure 5 and 6) as well as for other endpoints such as the release of inflammatory markers, CYP1A1/1B1 activity, cytotoxicity (see figure 7) and morphological changes (see figure 4).
- A dose- and a time-dependent effect was also observed after THS2.2 exposure but its overall impact on nasal Mucilair™ tissue cultures was always lower compared to 3R4F exposure and closer to air-exposed tissue cultures.

REFERENCES

1. **A network-based approach to quantifying the impact of biologically active substances.** *J. Hoeng, et al.* Drug Discovery Today, 2012 May 1; 28(9):1193-201.
2. **Construction of a computable cell proliferation network for non-diseased lung tissue.** *J. Westra, et al.* BMC Systems Biology, 2011 Jul 5:105.
3. **Construction of a computable cellular stress network for non-diseased lung and cardiovascular tissue.** *W.K. Schlage, et al.* BMC Systems Biology, 2011 Oct 5:168.
4. **Construction of a Computable Network Model for DNA Damage, Cell Death, Autophagy, and Senescence.** *S. Gebel, et al.* Bioinformatics and Biology Insights 2013 7:97-117.
5. **A modular cell-type focused inflammatory process network model for non-diseased pulmonary tissue.** *J. Westra, et al.* Bioinformatics and Biology Insights 2013 Jun 20; 7:167-92.
6. **Assessment of network perturbation amplitude by applying high-throughput data to causal biological networks.** *F. Martin, et al.* BMC Systems Biology 2012, 6:54.
7. **Quantification of biological network perturbations for mechanistic insight and diagnostics using two-layer causal models assessment of biological impact using transcriptomic data and mechanistic network models.** *F. Martin, et al.* BMC Bioinformatics. 2014 Jul 11;15(1):238.

PMI RESEARCH & DEVELOPMENT

SOCIETY FOR RESEARCH ON NICOTINE AND TOBACCO

Philip Morris International Research & Development, Quai Jeanrenaud 5, 2000 Neuchâtel, Switzerland
T: +41 58 242 21 13, **F**: +41 58 242 28 11, **W**: www.pmi.com