Compilation of in vitro studies evaluating the impact of exposure to aerosols from heated tobacco products and an electronic vapor device on oral health and dental esthetics

Filippo Zanetti[1], Alexander K. Nussbaum[2], Anita Iskandar[1], Xiaoyi Zhao[3,4], Yanfang Ren[3], Shoaib Majeed[1], Nikolai V. Ivanov[1], Manuel C. Peitsch[1], Julia Hoeng[1]

[1]Philip Morris International R&D, Philip Morris Products S.A., Quai Jeanrenaud 5, CH-2000 Neuchâtel, Switzerland; [2]Philip Morris GmbH, Am Haag 14, 82166 Gräfeling, Germany; [3]University of Rochester Eastman Institute for Oral Health, Rochester, New York, USA; [4]Peking University School of Stomatology, Beijing, China

Introduction

Cigarette smoke (CS) causes serious diseases and has detrimental effects on human health, including oral health. Smoking can lead to gingivitis, periodontitis, tooth loss, and mouth cancer [1]. CS is also a risk factor for tooth discoloration. Pigmented compounds present in the particulate phase generated by combustion of tobacco may cause discoloration of dental hard tissues and restorative materials [2]. Switching from cigarette smoking to using reduced-risk products (RRP) has the potential to reduce the harm and dental esthetic concerns associated with smoking. However, rigorous scientific studies are necessary to demonstrate the potentially reduced detrimental effects of exposure to aerosols from RRPs relative to those of exposure to CS. In this poster, we summarize the results of numerous *in vitro* studies that investigated the impact of exposure to aerosols from different RRPs (2 heated tobacco products [HTP] and 1 electronic vapor [e-vapor] product) on oral cell cultures and dental coloration. In addition, we present the results of a study aimed at characterizing the chemical compounds associated with tooth discoloration.

Study designs

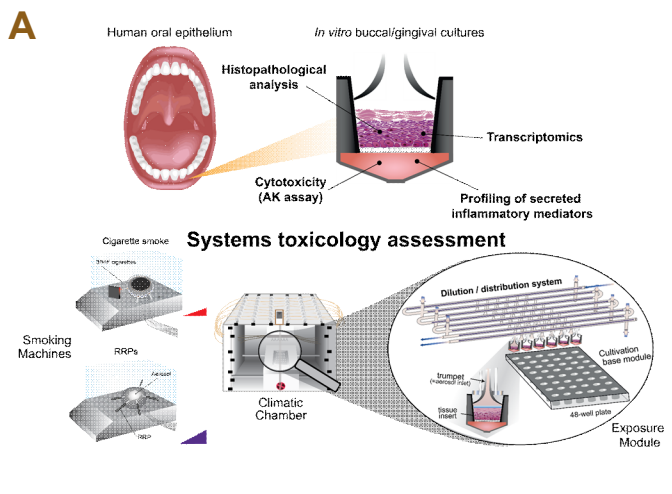
A
Human oral epithelium
In vitro buccal/gingival cultures
Histopathological analysis
Transcriptomics
Cytotoxicity (AK assay)
Profiling of secreted inflammatory mediators
Systems toxicology assessment
Cigarette smoke
Smoking Machines
RRPs
Dilution / distribution system
Climatic Chamber
Exposure Module

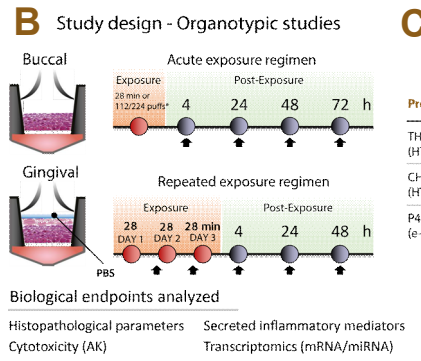
B Study design - Organotypic studies
Buccal
Acute exposure regimen
Exposure
Post-Exposure
4 24 48 72 h
Gingival
Repeated exposure regimen
28 DAY 1 28 DAY 2 28 min DAY 3
4 24 48 h
PBS
Biological endpoints analyzed
Histopathological parameters
Cytotoxicity (AK)
Secreted inflammatory mediators
Transcriptomics (mRNA/miRNA)

C

Product	Organotypic culture	Exposure regime	Reference
THS 2.2 (HTP)	Buccal	Acute	[3]
	Gingival	Repeated	[4]
CHTP 1.2 (HTP)	Buccal	Acute	[5]
	Gingival	Repeated	[5]
P4M3v1.0 (e-vapor)	Buccal	Acute	[6]

Figure 1. Experimental design for oral organotypic studies.

(A) Human oral (gingival and buccal) organotypic epithelial cultures were grown and differentiated on a permeable membrane, with cell culture medium located underneath. Organotypic cultures were exposed in the Vitrocell® 24/48 exposure system (Vitrocell Systems GmbH, Waldkirch, Germany). A climatic chamber contains an exposure module in which up to 48 wells can be exposed simultaneously to up to 8 dilutions of an aerosol/smoke. The cultures were exposed to CS or aerosol at concentrations matched at the level of nicotine deposited in the exposure module of the Vitrocell® exposure system.

(B) Study design — Endpoints and post-exposure time points. Buccal cultures were acutely (28 min) exposed, while gingival cultures were repeatedly exposed (28 min per day for 3 days) to diluted smoke from 3R4F reference cigarettes (University of Kentucky, Lexington, KY, USA), diluted/undiluted HTP aerosol, or 112/224 puffs of an e-vapor product aerosol (*). Exposure to air was used as control. Arrows indicate the collection time of the samples. PBS (100 µL) was placed on the apical side to mimic the moistening from saliva in gingival cultures. PBS, phosphate-buffered saline.

(C) Overview of the products investigated in the different studies. THS 2.2, Tobacco Heating System 2.2; CHTP 1.2, Carbon-Heated Tobacco Product 1.2; P4M3v1.0, e-vapor product using *MESH*™ technology (Philip Morris International, Neuchâtel, Switzerland).

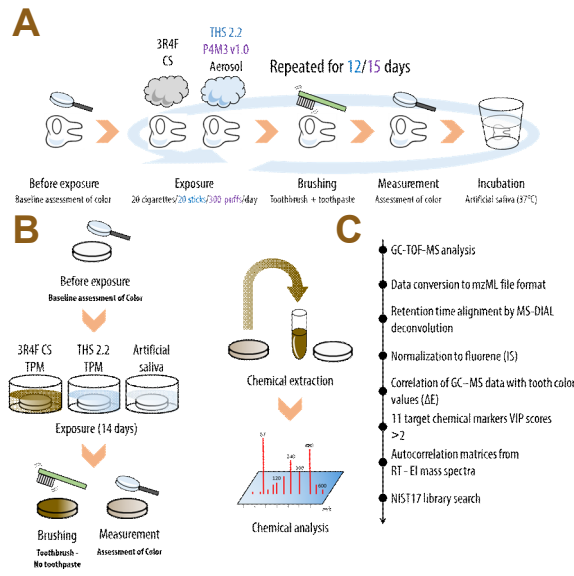
A
3R4F CS
THS 2.2 P4M3 v1.0 Aerosol
Repeated for 12/15 days
Before exposure
Exposure
Brushing
Measurement
Incubation
B
Before exposure
3R4F CS TPM
THS 2.2 TPM
Artificial saliva
Exposure (14 days)
Brushing
Measurement
C
Chemical extraction
Chemical analysis
GC-TOF-MS analysis
Data conversion to mzML file format
Retention time alignment by MS-DIAL deconvolution
Normalization to fluorene (IS)
Correlation of GC–MS data with tooth color values (ΔE)
11 target chemical markers VIP scores >2
Autocorrelation matrices from RT - EI mass spectra
NIST17 library search

Figure 2. Dental discoloration studies and determination of chemical compounds associated with tooth discoloration.

(A) Human premolar teeth restored with a composite resin filling (Filtek™ Supreme Ultra, M ESPE, St. Paul, MN, USA) were exposed in the Vitrocell® 24/48 exposure system to undiluted 3R4F CS or THS 2.2 aerosol from 20 cigarettes or tobacco sticks, respectively, [7] or to P4M3 v1.0 aerosols (300 puffs per day) [8] for 12/15 days. Exposure to different beverages (coffee, red wine, and soy sauce) for 56 min per day was also included in [8]. The teeth were brushed with a standard toothbrush and regular toothpaste every week (THS 2.2 study) or every day (P4M3 v1.0 study). Color was assessed every 4 days (THS 2.2 study) or 5 days (P4M3 v1.0 study).

(B) Bovine enamel discs were exposed to total particulate matter (TPM) from 3R4F CS, THS 2.2 aerosol, or artificial saliva with 2% ethanol (AS; control) continuously for 7 days under gentle agitation. Color was assessed at baseline and before and after brushing with a standard toothbrush without toothpaste. This procedure was repeated twice (total of 14 days of exposure). Samples were analyzed by GC–TOF-MS for compound identification in accordance with the workflow (C) [9]. EI, electron ionization; IS, internal standard; RT, retention time; VIP, variable importance in projection.

Selected results

Gingival THS 2.2 study [4] / Buccal P4M3 v1.0 study [6]

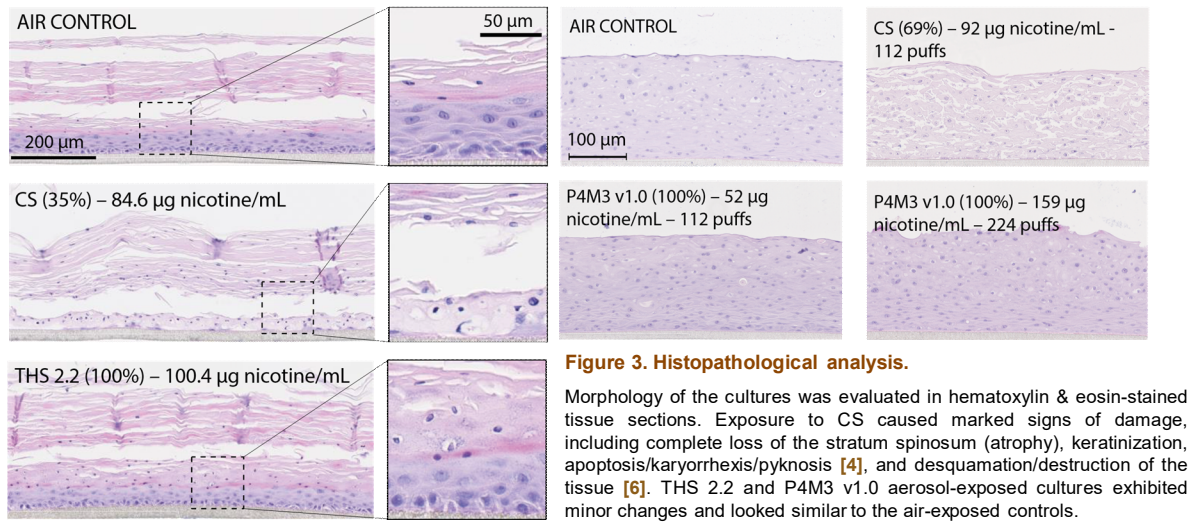
AIR CONTROL
50 µm
200 µm
CS (35%) – 84.6 µg nicotine/mL
THS 2.2 (100%) – 100.4 µg nicotine/mL
AIR CONTROL
100 µm
CS (69%) – 92 µg nicotine/mL - 112 puffs
P4M3 v1.0 (100%) – 52 µg nicotine/mL – 112 puffs
P4M3 v1.0 (100%) – 159 µg nicotine/mL – 224 puffs

Figure 3. Histopathological analysis.

Morphology of the cultures was evaluated in hematoxylin & eosin-stained tissue sections. Exposure to CS caused marked signs of damage, including complete loss of the stratum spinosum (atrophy), keratinization, apoptosis/karyorrhexis/pyknosis [4], and desquamation/destruction of the tissue [6]. THS 2.2 and P4M3 v1.0 aerosol-exposed cultures exhibited minor changes and looked similar to the air-exposed controls.

Gingival THS 2.2 study [4]

Fold- Changes (min – 1.0 – max)

After III exposure

	CS 49.4	CS 84.6	THS2.2 54.6	THS2.2 100.4
VEGFA	0.7*	0.5*	0.8*	0.9
TNFA	5.9*	3.8*	-	-
CCL5	0.5*	0.4*	0.5*	0.6*
MMP-9	0.5*	0.5*	0.8	0.8
MMP-1	7.5*	8.3*	1.8*	2.5*
CCL2	-	1.4	0.8	1.2
IP-10	1.4	1.4	0.9	1.1
IL-8	4.1*	3.7*	-	1.2
IL-6	0.6	0.5*	0.7	0.8
IL-1B	0.9	1.2	-	0.8
IL-1A	11*	16*	1.1	1.3
CXCL1	2*	2.2*	0.9	-
CSF2	14*	11*	1.5	1.8*
CSF3	4.4*	4.6*	1.4	1.5*

Buccal CHTP 1.2 study [5]

72 h Post-Exposure

µg nicotine/mL	CS 0.46	CS 0.82	CHTP1.1 0.45	CHTP1.1 0.96	CHTP1.1 1.52
VEGFA	0.9	0.8*	0.9	1.0	1.1
TNFA	-	-	-	-	-
CCL5	-	-	-	-	-
MMP-9	-	-	-	-	-
MMP-1	3.0*	5.6*	1.3*	1.6*	1.7*
CCL2	-	-	-	-	-
IP-10	0.7*	0.9	0.7*	0.6*	0.7*
IL-8	0.9	0.9	0.8*	0.8*	0.8*
IL-6	0.4*	0.2*	0.7*	0.6*	0.6
IL-4	-	-	-	-	-
IL-1B	8.2*	12*	1.0	1.5	1.6*
IL-1A	1.2	1.5	0.8	0.9	0.9
CXCL1	-	-	-	-	-
CSF2	0.8*	1.0	0.8	0.8	0.8
CSF3	1.5	1.1	0.9	1.1	1.3
TGFA	3.1*	11*	0.9*	1.0	1.1

Figure 4. Analysis of inflammatory mediators secreted in the basolateral medium of organotypic cultures.

The concentrations of released inflammatory mediators were measured in the basolateral medium by using a Luminex®-based technology (Luminex, Austin, TX, USA). The fold changes are expressed relative to air-exposed controls. Red and blue shading indicates significant differences (increases and decreases, respectively) between the aerosol-exposed and air-exposed samples (*p < 0.05).

Gingival CHTP 1.2 study [5] / Buccal P4M3 v1.0 study [6]

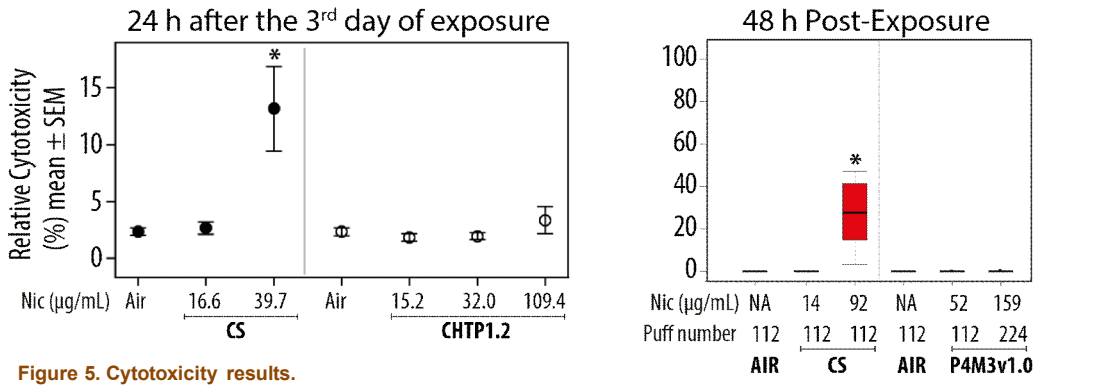
24 h after the 3rd day of exposure
Relative Cytotoxicity (%) mean ± SEM
Nic (µg/mL) Air 16.6 39.7 Air 15.2 32.0 109.4
CS
CHTP1.2
48 h Post-Exposure
Nic (µg/mL) NA 14 92 NA 52 159
Puff number 112 112 112 112 112 224
AIR CS AIR P4M3v1.0

Figure 5. Cytotoxicity results.

The activity of adenylate kinase (AK) was measured in the basolateral medium by using the ToxiLight™ BioAssay kit (Lonza, Rockland, MA, USA). CS-exposed cultures exhibited increased cytotoxicity relative to air controls. Cytotoxicity was not observed following CHTP 1.2 or P4M3 v1.0 aerosol exposures (*p < 0.05).

Gingival CHTP 1.2 study [5] / Buccal P4M3 v1.0 study [6]

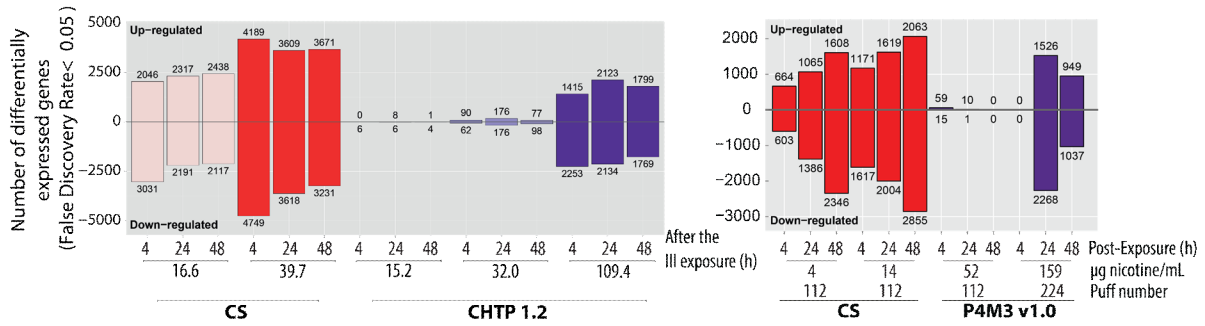
Number of differentially expressed genes (False Discovery Rate< 0.05)
Up-regulated
Down-regulated
After the III exposure (h)
16.6 39.7 15.2 32.0 109.4
CS
CHTP 1.2
Post-Exposure (h)
µg nicotine/mL
Puff number
4 14 52 159
112 112 112 224
CS
P4M3 v1.0

Figure 6. Gene expression changes.

Analysis of gene expression was performed by using the Affymetrix GeneChip® technology (Affymetrix, Santa Clara, CA, USA). The bar plots represent the numbers of significantly differentially expressed genes (upregulated and downregulated mRNAs) in buccal and gingival cultures at 4, 24, and 48 h after exposure (3rd day of exposure in case of the gingival CHTP 1.2 study) to CS and P4M3 v1.0 or CHTP 1.2 aerosols, respectively, relative to the expression levels in the air-exposed cultures (FDR < 0.05).

THS 2.2 dental study [7] / Dental study P4M3 v1.0 [8]

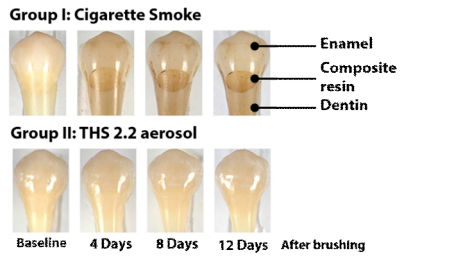
Group I: Cigarette Smoke
Enamel
Composite resin
Dentin
Group II: THS 2.2 aerosol
Baseline
4 Days
8 Days
12 Days
After brushing

	Enamel 4 Days	Enamel 8 Days	Enamel 12 Days	Dentin 4 Days	Dentin 8 Days	Dentin 12 Days	Composite resin 4 Days	Composite resin 8 Days	Composite resin 12 Days
CS	5.2 ± 2.2	7.1 ± 2.3	8.8 ± 2.6	13.9 ± 3.0	18.2 ± 3.9	21.3 ± 4.4	12.5 ± 2.6	19.0 ± 4.3	25.6 ± 3.8
THS 2.2	1.2 ± 0.8	2.0 ± 1.0	2.8 ± 1.2	1.2 ± 0.6	2.4 ± 0.7	3.1 ± 0.8	1.1 ± 0.9	1.9 ± 0.8	3.0 ± 1.0
p*	<0.0001	<0.0001	<0.0001	<0.0001	<0.0001	<0.0001	<0.0001	<0.0001	<0.0001

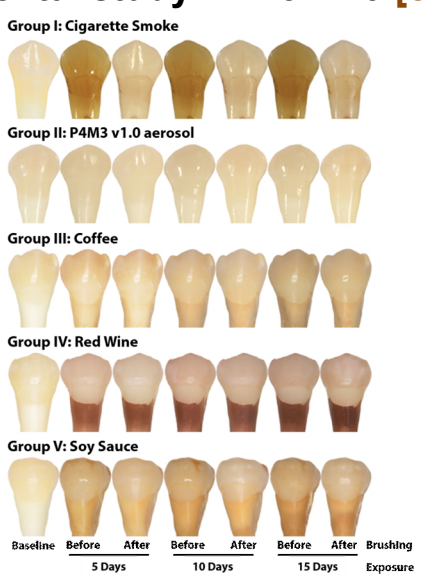
Group I: Cigarette Smoke
Group II: P4M3 v1.0 aerosol
Group III: Coffee
Group IV: Red Wine
Group V: Soy Sauce
Baseline
Before After
5 Days
Before After
10 Days
Before After
15 Days
Brushing
Exposure

Figure 7. Dental discoloration.

The color of the teeth and enamel blocks was assessed in the Commission Internationale de l'Eclairage L*a*b* (CIE Lab) color space by using an Olympus CrystalEye® dental spectrophotometer (Olympus, Tokyo, Japan). ΔE values (Table) indicate a difference in color from the baseline measurements.

Chemical characterization — THS 2.2 study [9]

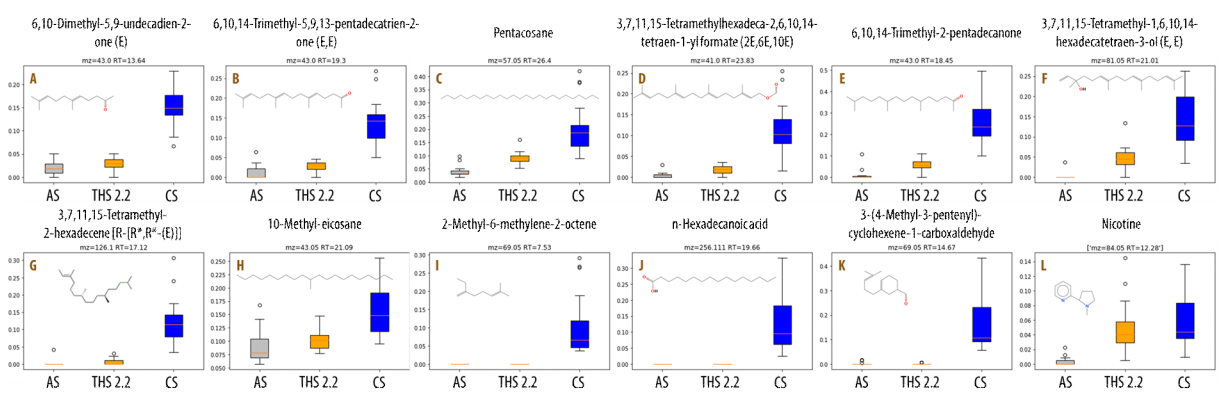
6,10-Dimethyl-5,9-undecadien-2-one (E)
6,10,14-Trimethyl-5,9,13-pentadecatrien-2-one (E,E)
Pentacosane
3,7,11,15-Tetramethylhexadeca-2,6,10,14-tetraen-1-yl formate (2E,6E,10E)
6,10,14-Trimethyl-2-pentadecanone
3,7,11,15-Tetramethyl-1,6,10,14-hexadecatetraen-3-ol (E, E)
3,7,11,15-Tetramethyl-2-hexadecene [R-[R*,R*-(E)]]
10-Methyl-eicosane
2-Methyl-6-methylene-2-octene
n-Hexadecanoic acid
3-(4-Methyl-3-pentenyl)-cyclohexene-1-carboxaldehyde
Nicotine
AS THS 2.2 CS

Figure 8. Box plots and chemical structures of the 11 compounds associated with tooth discoloration and nicotine.

(A-K) Eleven compounds were singled out as the main chemical contributors to the staining caused by CS TPM (blue boxplot) in bovine enamel discs, and most of these compounds were of terpenoid origin. Several of these compounds were also detected in THS 2.2 aerosol TPM-exposed enamel (orange boxplots), but at lower levels. The area ratio of the most intense fragment ion was normalized against m/z 166 of the spiked IS (fluorene). (L) Nicotine could not be identified as a significant source of discoloration.

Conclusions

Organotypic Studies

- A systems toxicology approach was applied for biological impact assessment of THS 2.2, CHTP 1.2, and P4M3 v1.1 aerosols relative to 3R4F CS in human organotypic oral epithelial cultures. Multiple endpoints were combined toward comprehensive assessment of the exposure effects.
- The results showed that cytotoxicity, morphological changes, inflammatory response profiles, and mRNA changes were less extensive and less pronounced in cultures exposed to the aerosols from the two HTP products and the e-vapor product when compared with the effects of CS exposure at similar nicotine concentrations.
- When similar changes were observed between samples exposed to RRPs aerosols and CS, these occurred upon exposure to higher concentrations of RRPs aerosols than CS (e.g., gene expression changes are similar between P4M3 v1.0 aerosol at 159 µg nicotine/mL and CS at 14 µg nicotine/mL).
- The biological alterations observed in oral cultures exposed to CS replicate the changes that occur in smokers (e.g., decreased IL-6 secretion, increased tissue keratinization) or in patients with gingival inflammation (e.g., increased secretion of MMP-1).

Dental Studies

- THS 2.2 aerosol discolored tooth enamel, dentin, and composite resins to a much lesser extent than 3R4F CS.
- Tooth discoloration associated with P4M3 v1.0 aerosol exposure was minimal and the lowest when compared with the discoloration induced by CS, red wine, coffee, and soy sauce.
- Regular tooth brushing affects the outcomes of different exposures on different substrates, including enamel, dentin, and composite resin restorations.
- Eleven compounds were singled out as the main chemical contributors to CS TPM staining, and most of them were terpenoids. They may have originated from the degradation products of tobacco constituents, tobacco flavors, or pyrolysis products of plant material.
- Nicotine could not be identified as a significant source of discoloration by our correlation analysis or univariate analyses.

References

[1] Reibel J. Tobacco and oral diseases. Update on the evidence, with recommendations. Med Princ Pract 2003;12 Suppl 1:22-32.

[2] Watts A, Addy M. Tooth discolouration and staining: a review of the literature. Br Dent J 2001;190(6):309-16.

[3] Zanetti F, Sewer A, Mathis C, et al. Systems Toxicology Assessment of the Biological Impact of a Candidate Modified Risk Tobacco Product on Human Organotypic Oral Epithelial Cultures. Chem Res Toxicol 2016;29(8):1252-69.

[4] Zanetti F, Titz B, Sewer A, et al. Comparative systems toxicology analysis of cigarette smoke and aerosol from a candidate modified risk tobacco product in organotypic human gingival epithelial cultures: A 3-day repeated exposure study. Food Chem Toxicol 2017;101:15-35.

[5] Zanetti F, Sewer A, Scotti E, et al. Assessment of the impact of aerosol from a potential modified risk tobacco product compared with cigarette smoke on human organotypic oral epithelial cultures under different exposure regimens. Food Chem Toxicol 2018;115:148-69.

[6] Iskandar AR, Zanetti F, Marescotti D, et al. Application of a multi-layer systems toxicology framework for in vitro assessment of the biological effects of Classic Tobacco e-liquid and its corresponding aerosol using an e-cigarette device with MESH technology. Arch Toxicol 2019;93(11):3229-47.

[7] Zanetti F, Zhao X, Pan J, et al. Effects of cigarette smoke and tobacco heating aerosol on color stability of dental enamel, dentin, and composite resin restorations. Quintessence Int 2019;50(2):156-66.

[8] Zhao X, Zanetti F, Wang L, et al. Effects of different discoloration challenges and whitening treatments on dental hard tissues and composite resin restorations. J Dent 2019;89:103182.

[9] Haiduc A, Zanetti F, Zhao X, et al. Analysis of chemical deposits on tooth enamel exposed to total particulate matter from cigarette smoke and tobacco heating system 2.2 aerosol by novel GC-MS deconvolution procedures. J Chromatogr B Analyt Technol Biomed Life Sci 2020;1152:122228.

All raw data from the studies presented in this poster are available at www.Intervals.com.

Deutsche Gesellschaft für Parodontologie, November 13-14-2020

Competing Financial Interest: The research described in this poster was sponsored by Philip Morris International