Untargeted Metabolomics Application to Monitor Urine Samples by Ultra-High-Pressure Liquid Chromatography Coupled to High-Resolution Mass Spectrometry

Cs. Laszlo[1*], A. Haiduc[1], M. Fatarova[1], E. Dossin[1], F. de Jong[2], C. Beecher[2], A. Kamleh[3], P.A. Guy[1], N.V. Ivanov[1], M. Peitsch[1]
[1]PMI R&D, Philip Morris Products S.A., Quai Jeanrenaud 5, 2000 Neuchâtel, Switzerland; [2] IROA Technologies, Bolton, MA, USA; [3] ThermoFisher Scientific, Waltham, MA, USA
* Corresponding author

Objectives

- In order to meet the demands of high-throughput metabolomics, we established an efficient, easily transferable LC-HR-MS method for urine sample analysis.
- We employed the latest model of Vanquish tandem LC system (ThermoFisher) with considerable time saving benefit.
- Retention time (RT) drift across the columns was accounted for and RTs were re-aligned using dynamic time warping (DTW) [1,2].
- In order to improve the normalization strategy and implicitly obtain a more robust method for metabolite identification we have assessed the benefits of spiking urine samples with ^{13}C yeast extract (TruQuant IQQ Workflow kit, IROA) [3].
- Urine metabolite concentrations vary widely (up to 15-fold) due to water uptake [4] , as part of our normalization strategy, here we asses the importance of urine volume normalization based on creatinine levels using matrix matched calibration curve approach [5].

Methods

- Pooled human urine sample was thawed and centrifuged at 15,000 g for 15 minutes at 4 °C.
- Separation of metabolites was performed using two Hypersil Gold C18 columns (150 x 2.1 mm, 1.9 µm) operating in parallel and running with a fast linear gradient of acetonitrile containing 0.1% formic acid ramping from 5% to 95% in 15 min, alternating between the two columns at 0.5 ml/min. Column was kept at 50 °C.
- Dual-column operation improves the chromatography repeatability, maintains full occupancy of the MS instrument, enabling approx. 100 injections per day.
- Same urine sample (2 µl) was injected 80 times on each column in positive, and repeated in negative ESI mode.

Figure 1: Tandem operation of chromatographic columns: MS acquisition of column A while re-equilibration of column B . (100 h instrument time saved for 200 injections (pos/neg).

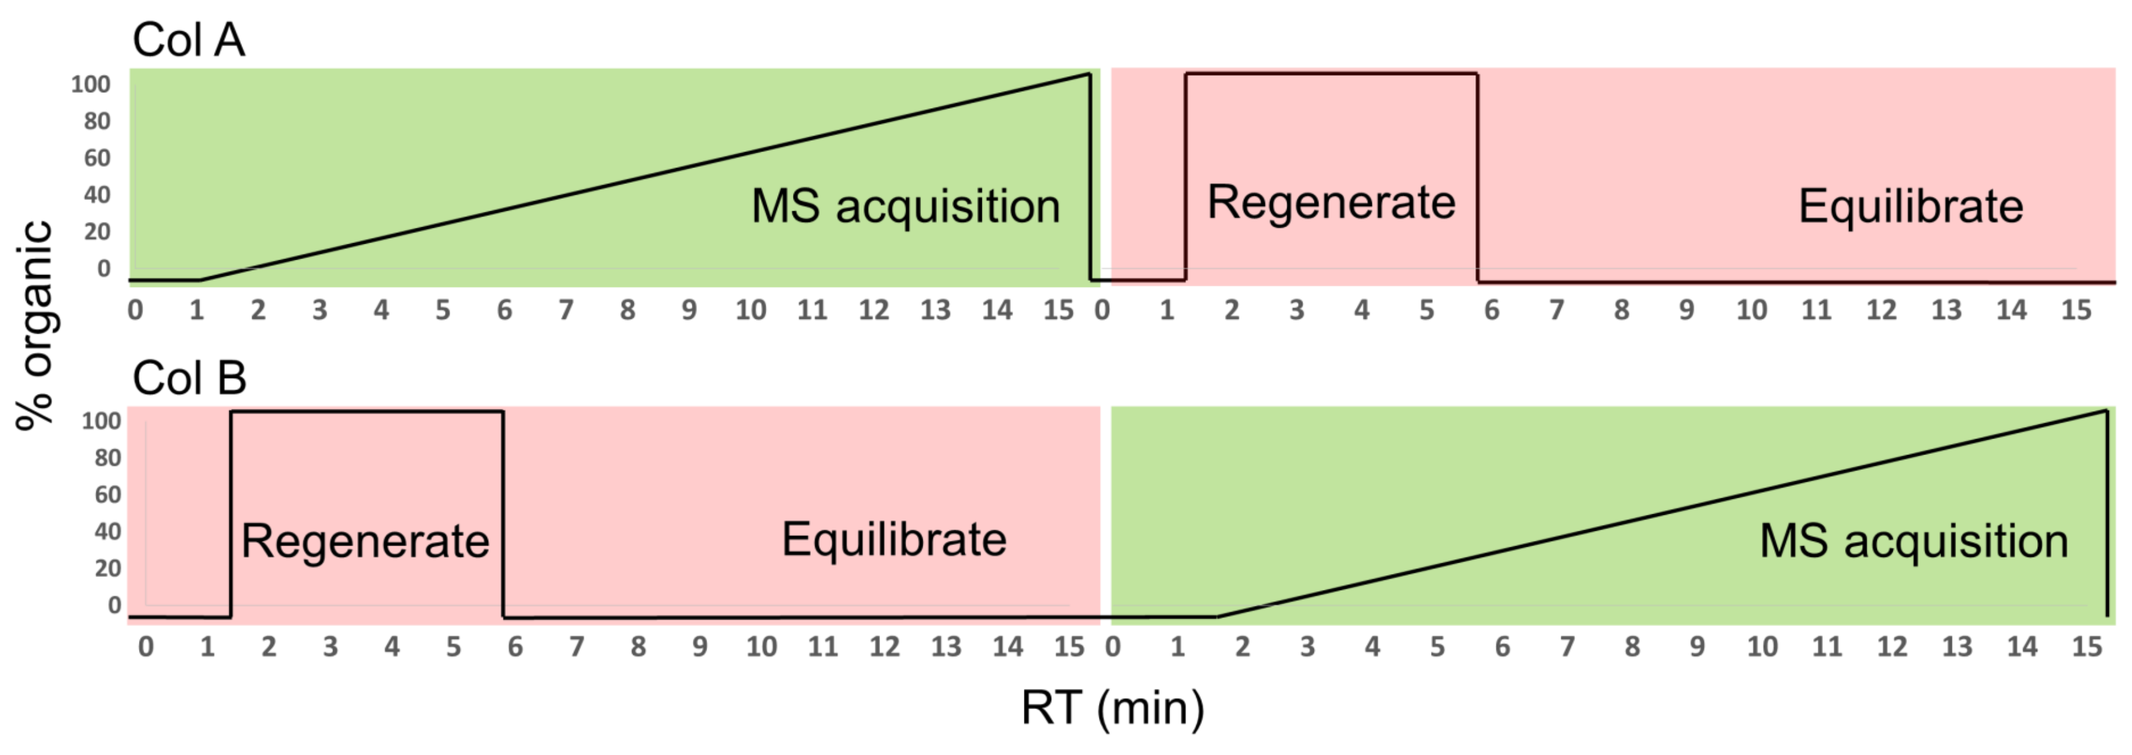
Col A
MS acquisition
Regenerate
Equilibrate
% organic
Col B
Regenerate
Equilibrate
MS acquisition
RT (min)

- Mass spectrometric detection was done on a Q Exactive HF mass spectrometer operating in positive and negative electrospray ionization FS acquisition modes with a mass range *m/z* 70-1050 and resolution 120k FWHM @ 200 *m/z*. Presented data is in positive mode unless otherwise stated.

Results

Retention time re-alignment across different columns or systems

- Dynamic time warping (DTW) is an algorithm for measuring similarity between two temporal sequences. In chromatography, DTW is using the Euclidean distance to construct the shortest path between a reference chromatogram and a target chromatogram that minimizes the sum of absolute differences. DTW aligns each point of the query chromatogram.

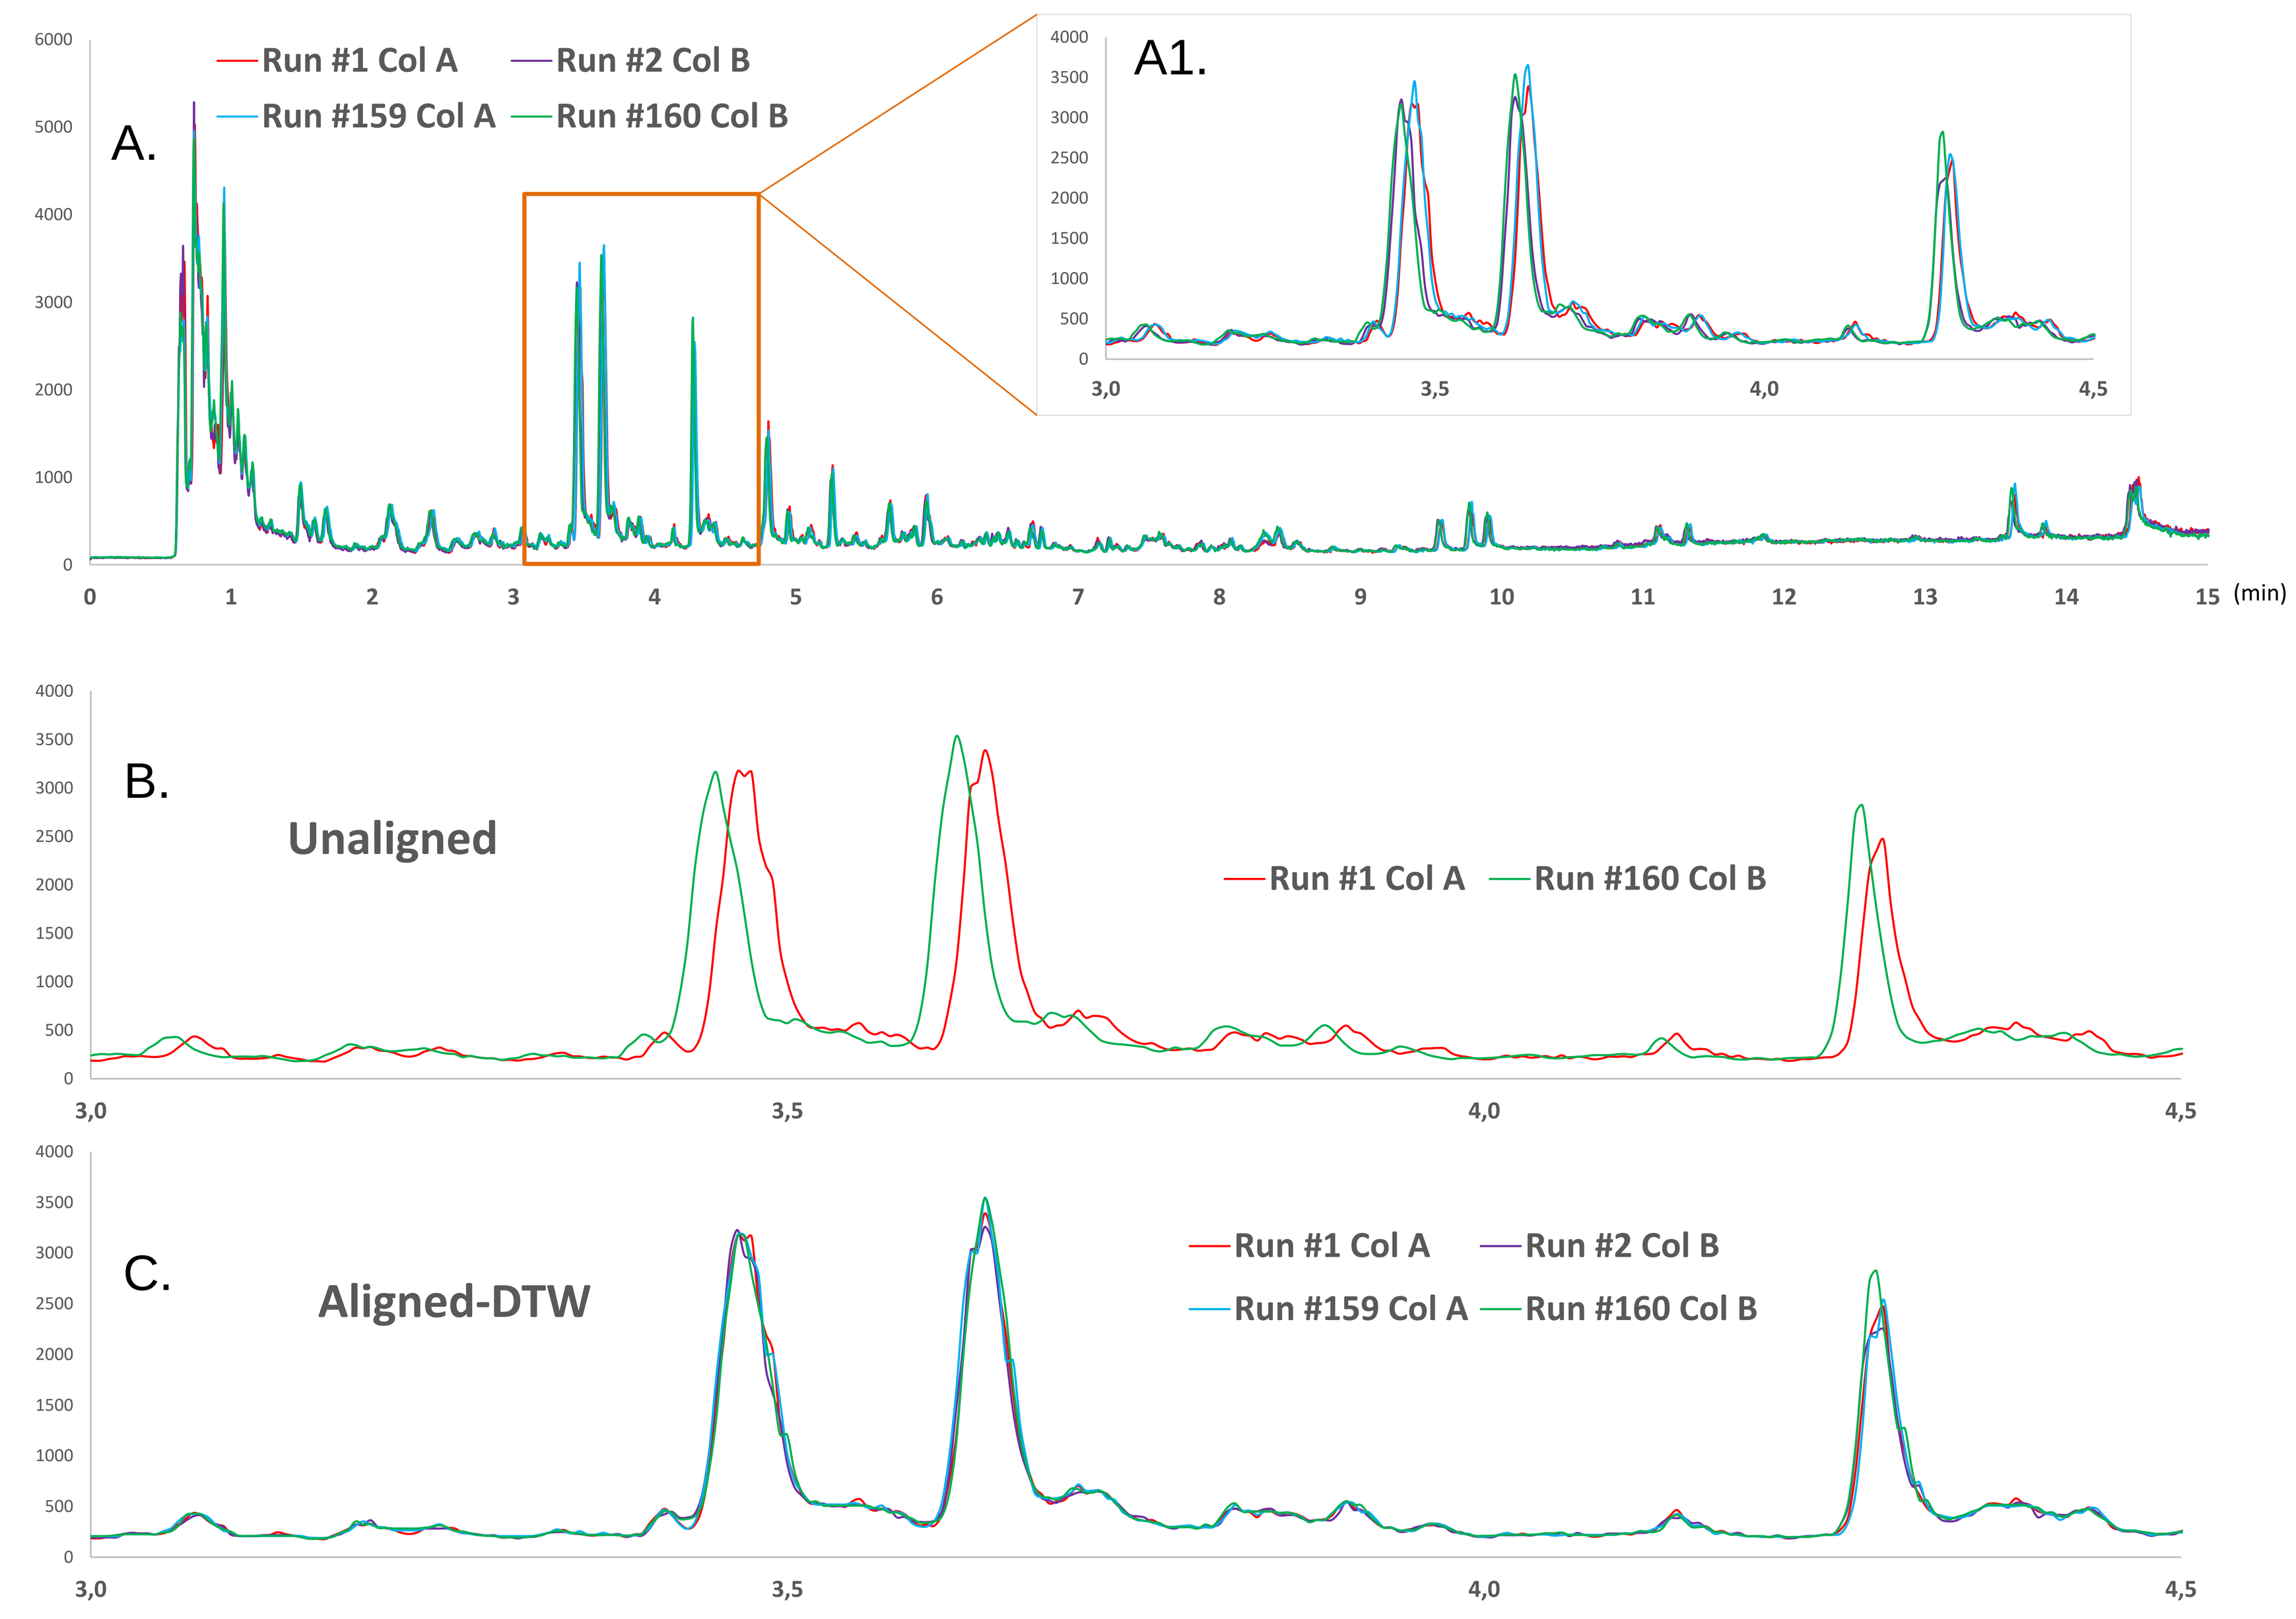
A.
Run #1 Col A
Run #2 Col B
Run #159 Col A
Run #160 Col B
A1.
(min)
B.
Unaligned
Run #1 Col A
Run #160 Col B
C.
Aligned-DTW
Run #1 Col A
Run #2 Col B
Run #159 Col A
Run #160 Col B

Figure 2: TIC of: (A) four injections on two columns, first and last injections on columns A and B, (A1) insert zoomed in from 3 to 4.5 min; (B) Unaligned injections on the two columns, (C) Aligned using DTW algorithm.

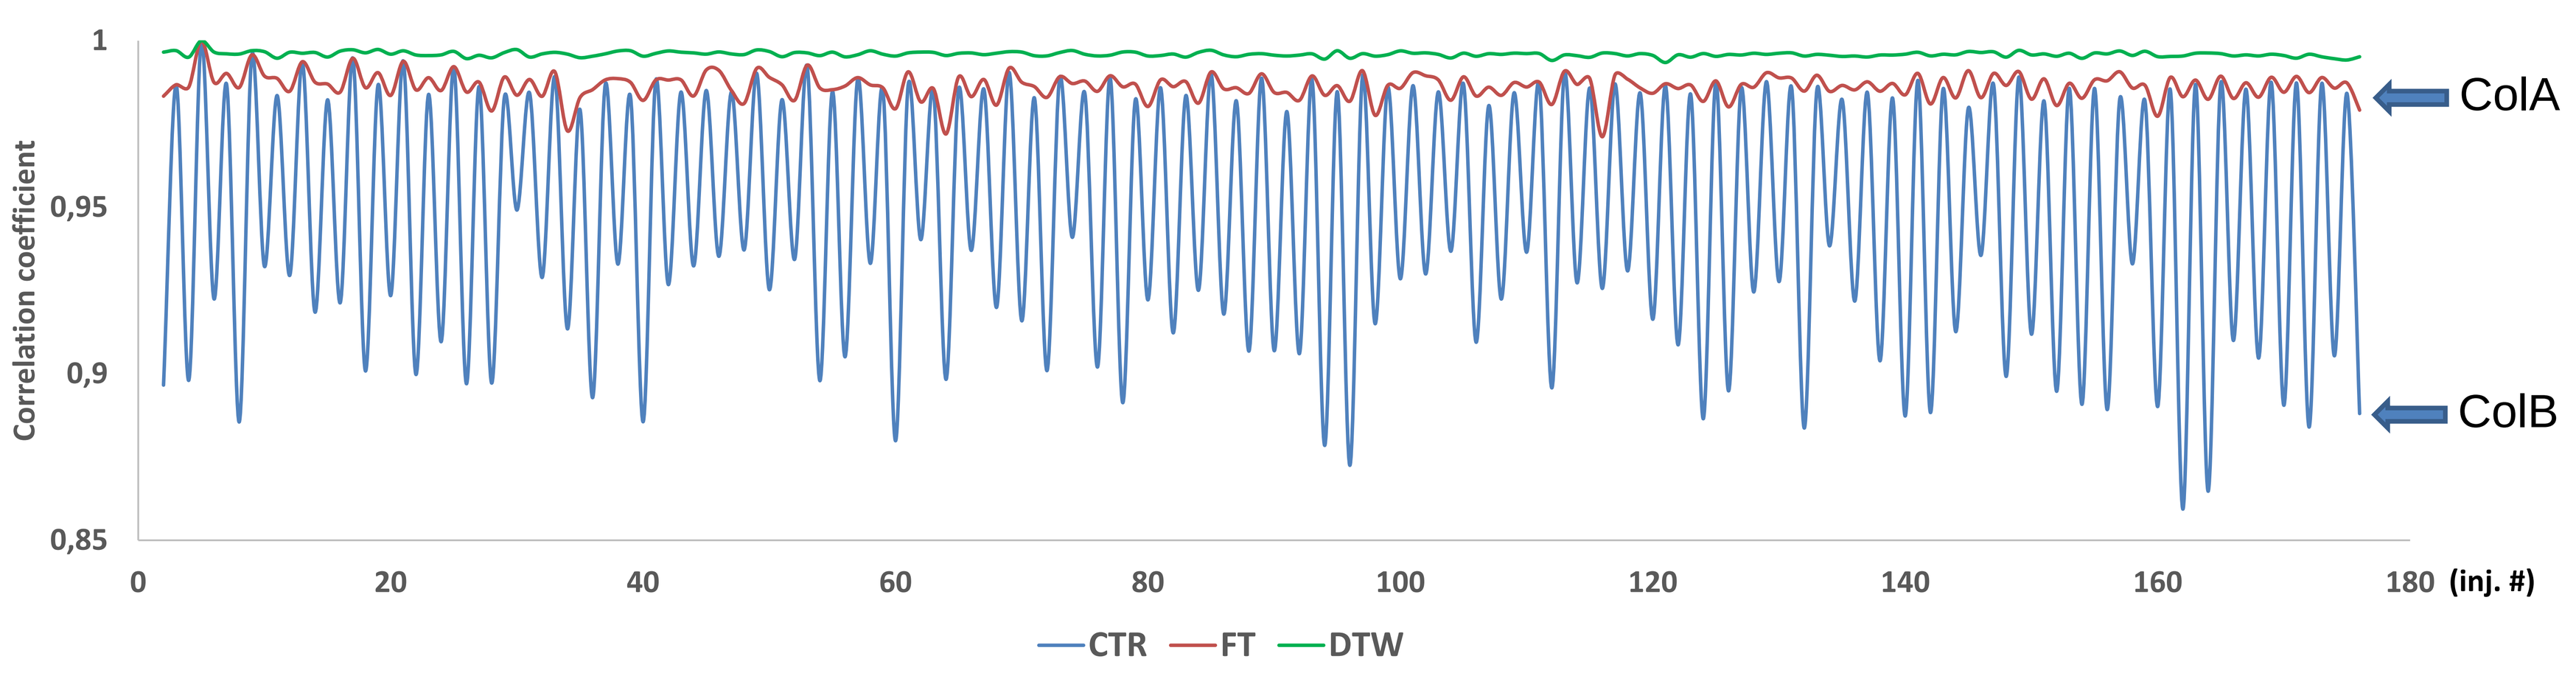
Correlation coefficient
ColA
ColB
(inj. #)
CTR
FT
DTW

Figure 3: Inter-sample correlation of total ion current (TIC), unaligned (blue), aligned with Fourier Transform (FT) (red) and aligned with DTW (green)

A.

	m/z166.08626		m/z120.08130			
	Phe_quan		Phe_qual		Ratio	
	Area	Height	Area	Height	Area	Height
CV (%)	9,9	9,7	8,8	8,6	3,7	4,9

B.

	DTW alignment			
	Avg. Area		RT (7.6min)	
	NO	YES	NO	YES
CV (%)	7,7	2,85	0,21	0,04

Table 1: A:Coefficient of variation (CV) of peak area and height of Phenylalanine qualifier and quantifier ions within 200 injections on both ColA and ColB; B: Average area CV% entire chromatogram and RT CV% at 7,6 min before and after DTW, 200 injections.

IROA ^{13}C yeast extract application for untargeted urine metabolomics

- Here we explore the possibilities offered by spiking pooled urine sample with a complex internal standard (IS) obtained from metabolically labeled yeast extract that contains hundreds of ^{13}C metabolites (IROA).

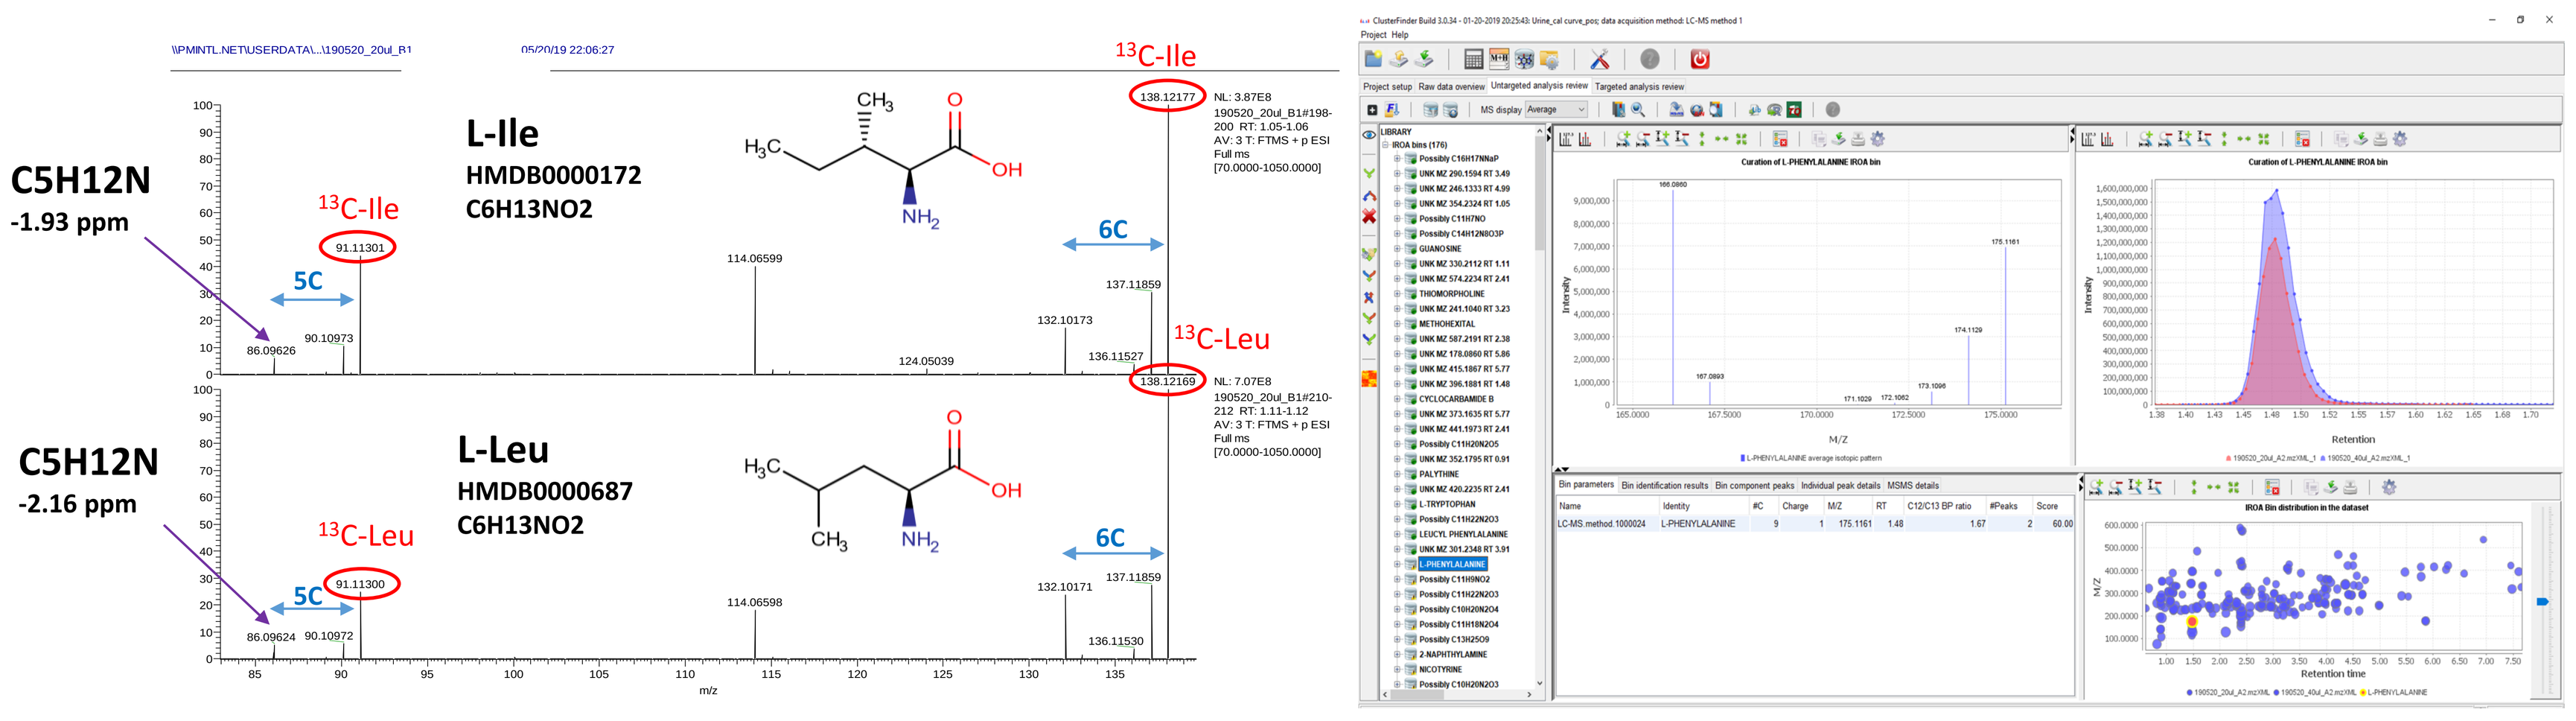
C5H12N
-1.93 ppm
L-Ile
HMDB0000172
C6H13NO2
13C-Ile
5C
6C
C5H12N
-2.16 ppm
L-Leu
HMDB0000687
C6H13NO2
13C-Leu

Figure 4: Left: example of ^{12}C and ^{13}C labelled Leucine (Leu) and Isoleucine (Ile) detection principle with the IROA, right: ClusterFinder analysis software screenshot.

- In order to build a calibration curve, 0 to 100 µl volume of urine were dried down under nitrogen flow and re-suspended in 40 µl of ^{13}C labelled metabolite solution as per fabricant recommendation [3].

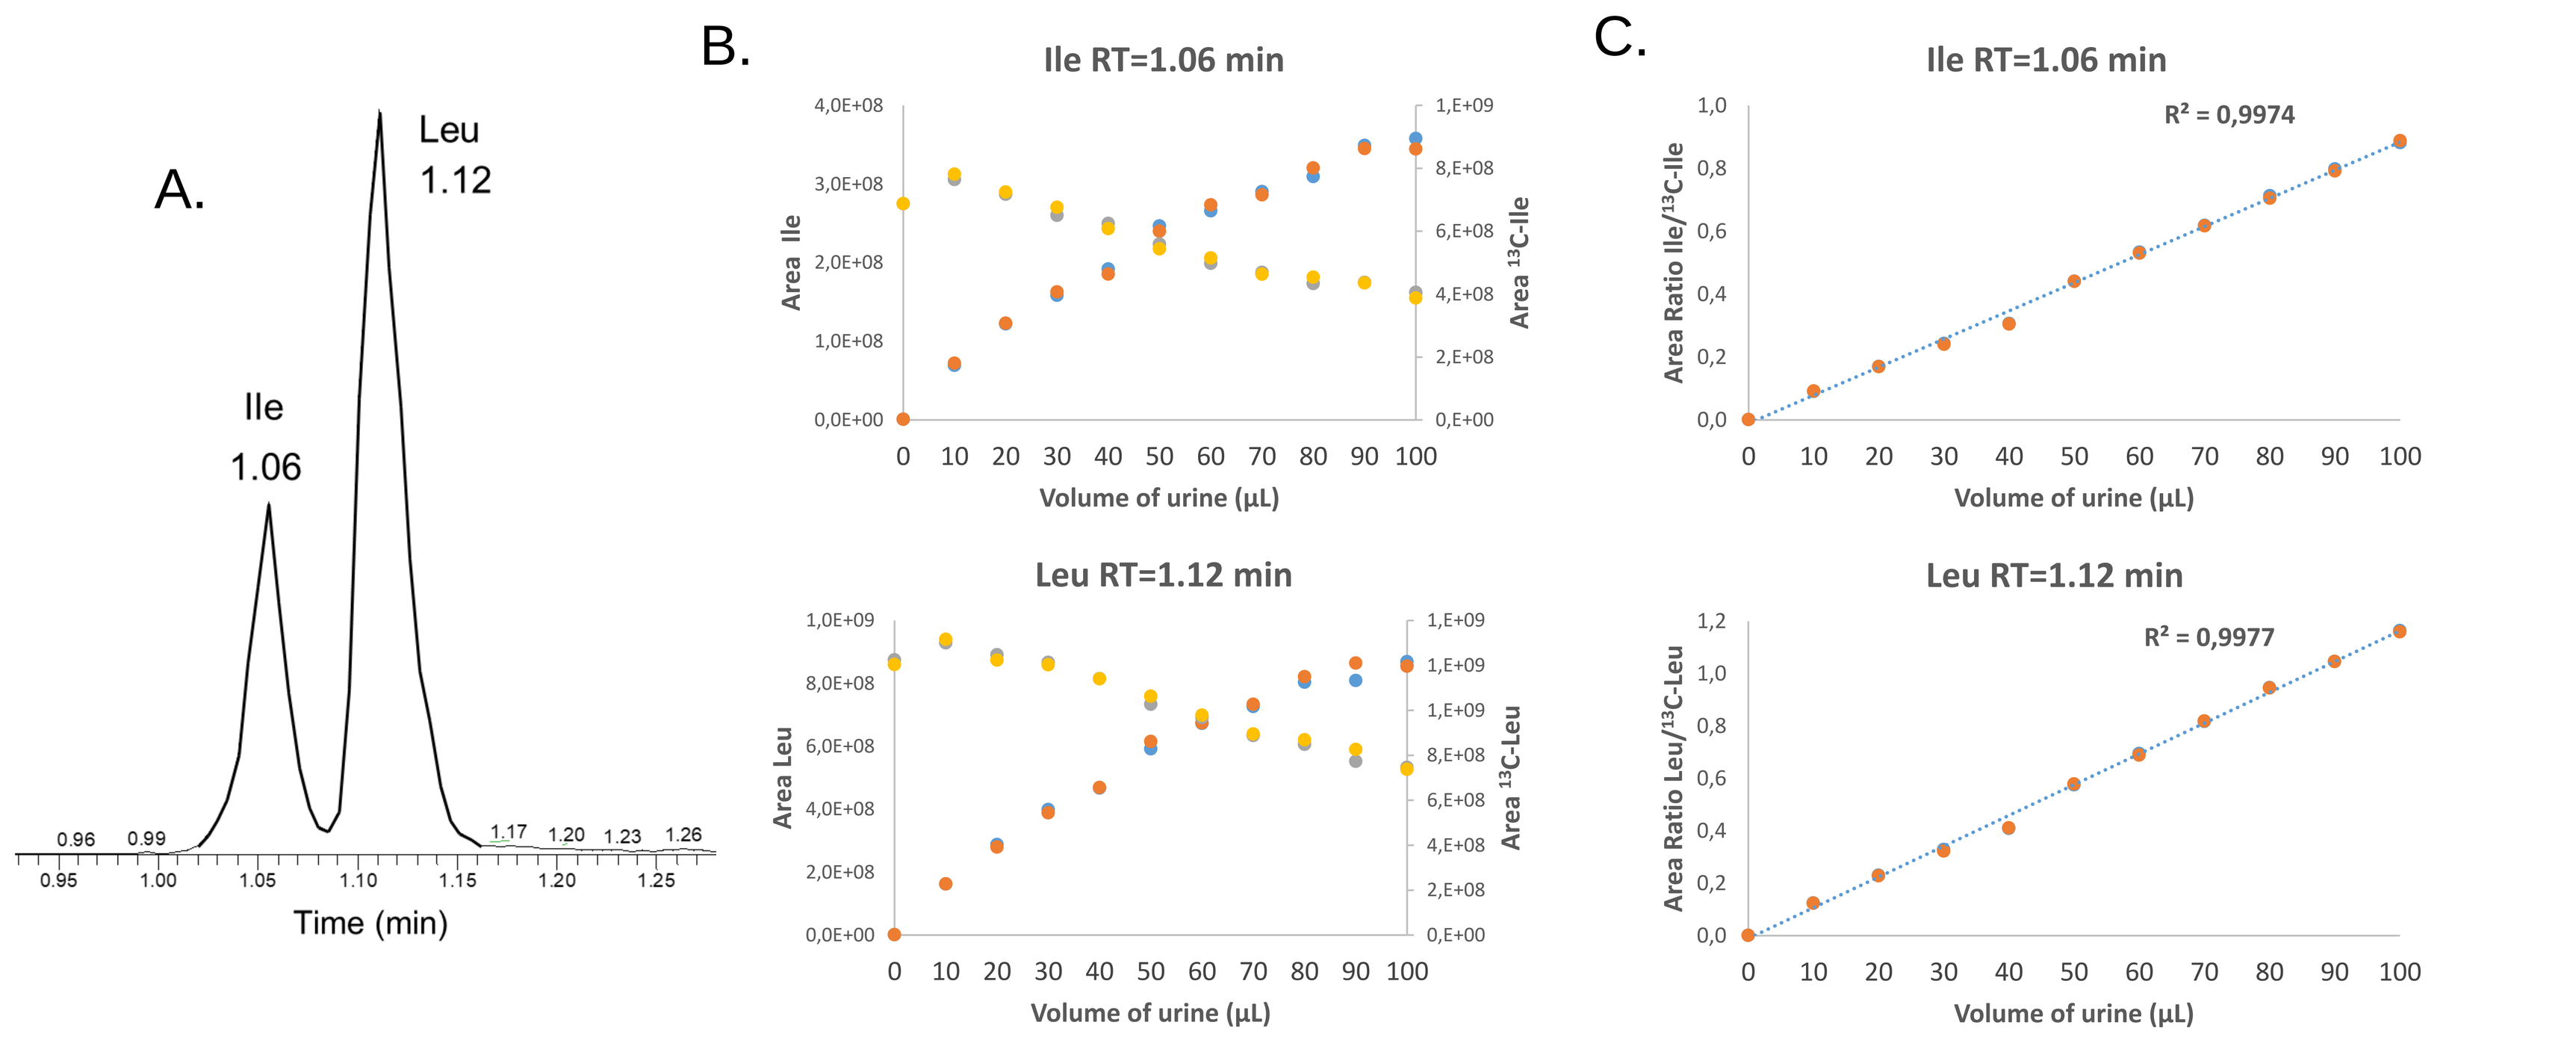
A.
Leu
1.12
Ile
1.06
Time (min)
B.
Ile RT=1.06 min
Area Ile
Area 13C-Ile
Volume of urine (µL)
Leu RT=1.12 min
Area Leu
Area 13C-Leu
C.
Ile RT=1.06 min
R² = 0,9974
Area Ratio Ile/13C-Ile
Leu RT=1.12 min
R² = 0,9977
Area Ratio Leu/13C-Leu

Figure 5: A: Leucine (Leu), Isoleucine (Ile) chromatographic separation; B: Leu and Ile peak areas determined by red dots, Secondary axis (right) and yellow dots represent areas corresponding to ions of the ^{13}C *internal standards. C: Leu and Ile normalized calibration curves. Linear regression R^2 values presented on graphs.*

Urine sample volume normalization

- Volume normalization consideration takes into account detected number of features as a function of measured creatinine levels, as well as ion suppression observed in ESI-MS

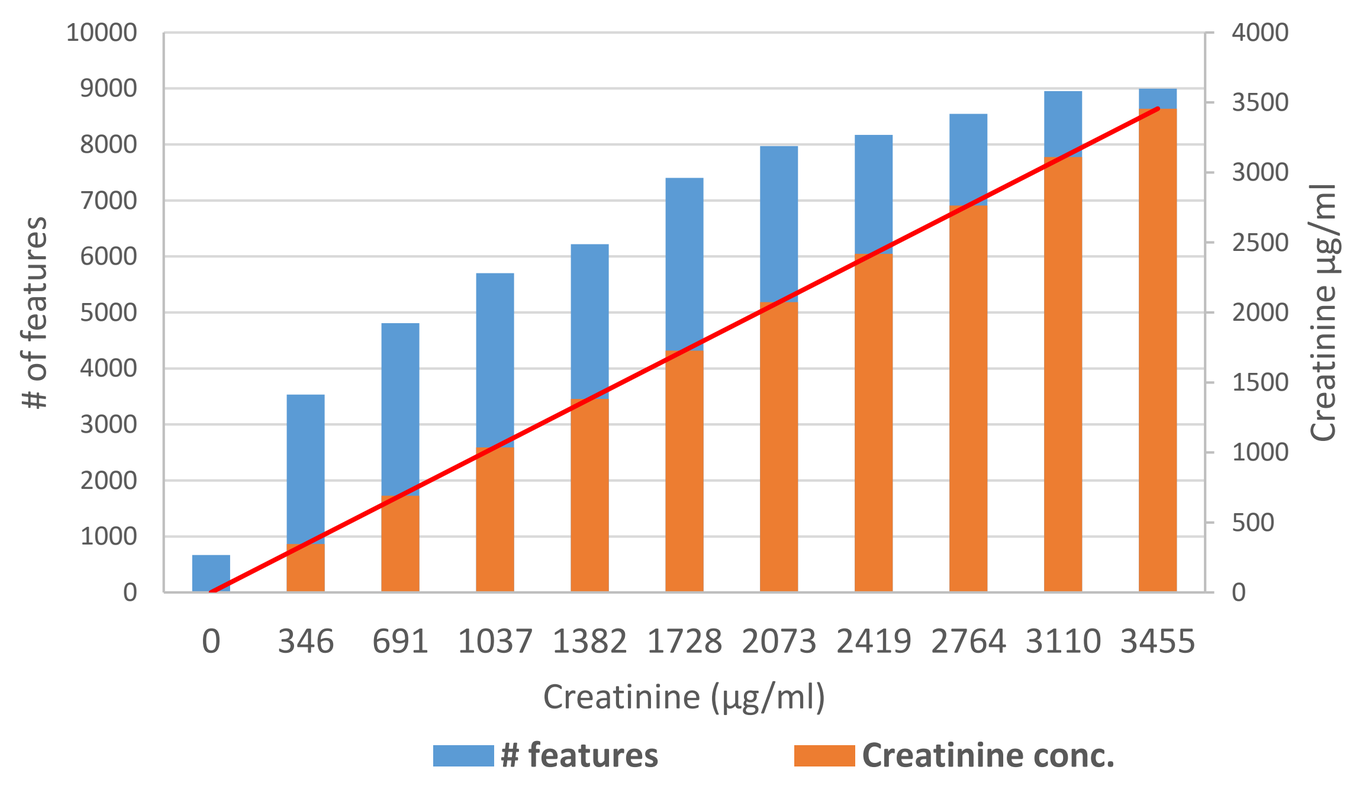
of features
Creatinine µg/ml
Creatinine (µg/ml)
features
Creatinine conc.

Creatinine (µg/ml)	# features	% gain
346	3535	NA
691	4810	36%
1037	5701	19%
1382	6218	9%
1728	7402	19%
2073	7970	8%
2419	8170	3%
2764	8545	5%
3110	8953	5%
3455	8996	0%

Figure 6: Number of total features detected (blue bars) by Compound Discoverer 3.0 (ThermoFisher) for increasing creatinine concentration levels. Values in table on the right show % gain in number of features compared to the previous creatinine concentration level.

- Ion suppression is a poorly understood phenomena consisting of the loss of ionization of analytes. Stable isotope labeled IS spiking is considered to be one of the best approaches to compensate for ion suppression. Below we look at the percent ion suppression of ^{13}C labelled Tryptophan

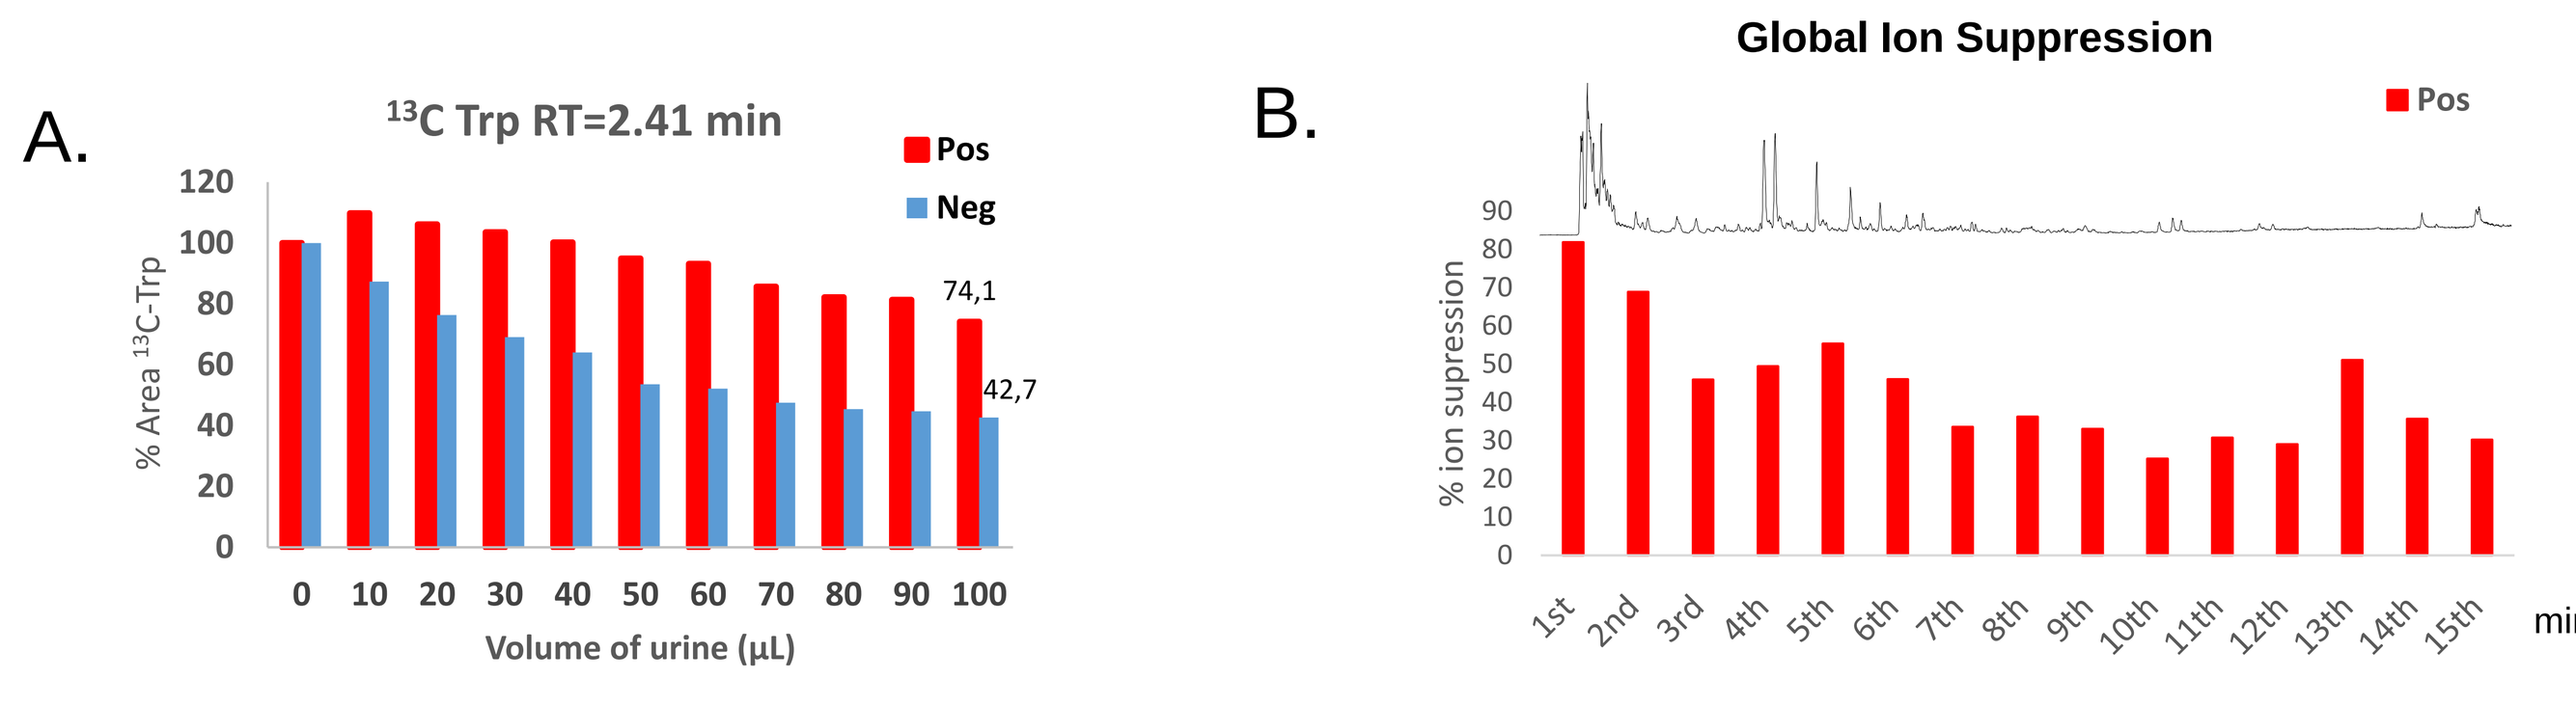
A.
13C Trp RT=2.41 min
Pos
Neg
% Area 13C-Trp
74,1
42,7
Volume of urine (µL)
B.
Global Ion Suppression
Pos
% ion supression
min

Figure 7: A. Percent ion suppression of ^{13}C Tryptophan IS in positive (red) and negative (blue) ionization modes. B. Global effect of ion suppression across the analysis with TIC insert overlap.

Conclusions

- ✓ We doubled the throughput due to dual column switching operating mode (~100 runs/day).
- ✓ DTW enables accurate peak alignment mandatory for metabolomics applications.
- ✓ Average CV of the area of all features was 2.85% from 200 injections of the same urine sample.
- ✓ ^{13}C IROA yeast extract proves to be beneficial for:
 - easier metabolite identification through better annotation (ClusterFinder software)
 - ion suppression monitoring and correction along the whole chromatogram
 - accurate normalization from associated ^{13}C proper IS or closely eluting alternative ^{13}C compounds
 - calibration curve building
- ✓ Indicative information was obtained regarding optimal urine creatinine concentration for sample volume normalization purposes.

References

[1] Clifford D. *et al*. **2009** *Anal. Chem*., 81, 3, 1000-1007

[2] Jiang W. *et al.* **2013** *Chromatographia* 76 1067–1078

[3] Beecher C. and de Jong FA. **2019** *Methods Mol Biol.* 1996 17-28

[4] Warrack B. *et al.* **2009** *J. Chromatogr. B*, 877, 547–552

[5] Kennedy A.D. *et al.* **2016** *Gen. Test and Mol. Biomarkers.* 20, *9, 485-495.*

Metabolomics 2019, June 23rd-27th, The Hague

Competing Financial Interest – The research described in this poster was sponsored by Philip Morris International