Systems toxicology assessment of a representative e-liquid formulation using human primary bronchial epithelial cells

Diego Marescotti, Carole Mathis, Vincenzo Belcastro, Patrice Leroy, Stefano Acali, Florian Martin, Rémi Dulize, David Bornand, Karine Baumer, Dariusz Peric, Emmanuel Guedj, Laura Ortega Torres, Matteo Biasioli, Matthieu Fuhrimann, Estella Fernandes, Felix Frauendorfer, Karsta Luettich, Ignacio Gonzalez Suarez, Davide Sciuscio, Nikolai V. Ivanov, Manuel C. Peitsch, and Julia Hoeng

Philip Morris International R&D, Philip Morris Products S.A., Quai Jeanrenaud 5, 2000 Neuchâtel, Switzerland

Introduction and objectives

Introduction: As a case study, we generated a representative flavor mixture by grouping a list of flavors commonly used in e-liquids based on common physicochemical properties and available toxicological data. Within each group, at least one representative flavor, predicted to show the highest potential toxicological effect, was selected to create a mixture, with 28 representative flavors dissolved in a Base Solution containing equivalent proportions of propylene glycol and vegetable glycerin and a total nicotine content of 0.6% (Table 1).

Objectives: A multi-step approach (Figure 1) was employed to evaluate:

1. The effects of exposing normal human bronchial epithelial cells (NHBE) in submerged condition to i) the flavor mixture and ii) corresponding 28 individual flavors.
2. The cytotoxic contribution of those flavors individually showing the highest cytotoxicity.

To achieve the latter, new mixtures were generated by selectively removing those flavors individually exhibiting the largest cytotoxic effects.

Table 1. Family, flavor name, and concentration in the e-liquid solution (in ppm and molarity) for the 28 flavor ingredients tested.

Group	Flavor name	Conc. ppm	Conc. molarity (mM)
XXII	2-ACETYLPYRIDINE	840	7.5
XXIII	2-ACETYLTHIAZOLE	16	0.2
IX	3-METHYL-2,4-NONANEDIONE	22	0.1
XV	3-(METHYLTHIO) PROPIONALDEHYDE	88	0.9
III	ALLYL HEXANOATE	280	1.6
XXV	ALPHA-PINENE	480	3.0
XVI	4-(PARA-HYDROXYPHENYL)-BUTAN-2-ONE	4800	29.2
I	BUTYRIC ACID	5600	61.3
VII	L-CARVONE	1200	7.7
XVII	CINNAMYL ALCOHOL	504	3.9
IV	CITRONELLOL	4800	26.3
I	ETHYL ACETATE	5600	57.3
I	ETHYL FORMATE	5600	69.6
X	ETHYL MALTOL	8160	58.2
XIX	2-ETHYL-3,6-DIMETHYLPYRAZINE	160	1.1
XIII	EUCALYPTOL	720	4.3
XIV	EUGENYL ACETATE	1440	7.5
XI	FURANEOL	1320	10.3
XX	GUAIACOL	107	1.0
V	HEPTAN-2-ONE	326	2.3
II	ISOBUTYL ALCOHOL	544	5.9
VI	LINALOOL	2400	13.5
VII	MENTHONE	1200	6.9
XXI	METHYL ANTHRANILATE	288	2.2
XXIV	CYCLOTENE	1756	15.7
XVIII	METHYL SALICYLATE	2320	17.9
XII	PHENETHYL ALCOHOL	2840	32.1
VIII	GAMMA-VALEROLACTONE	3000	31.5

Methods

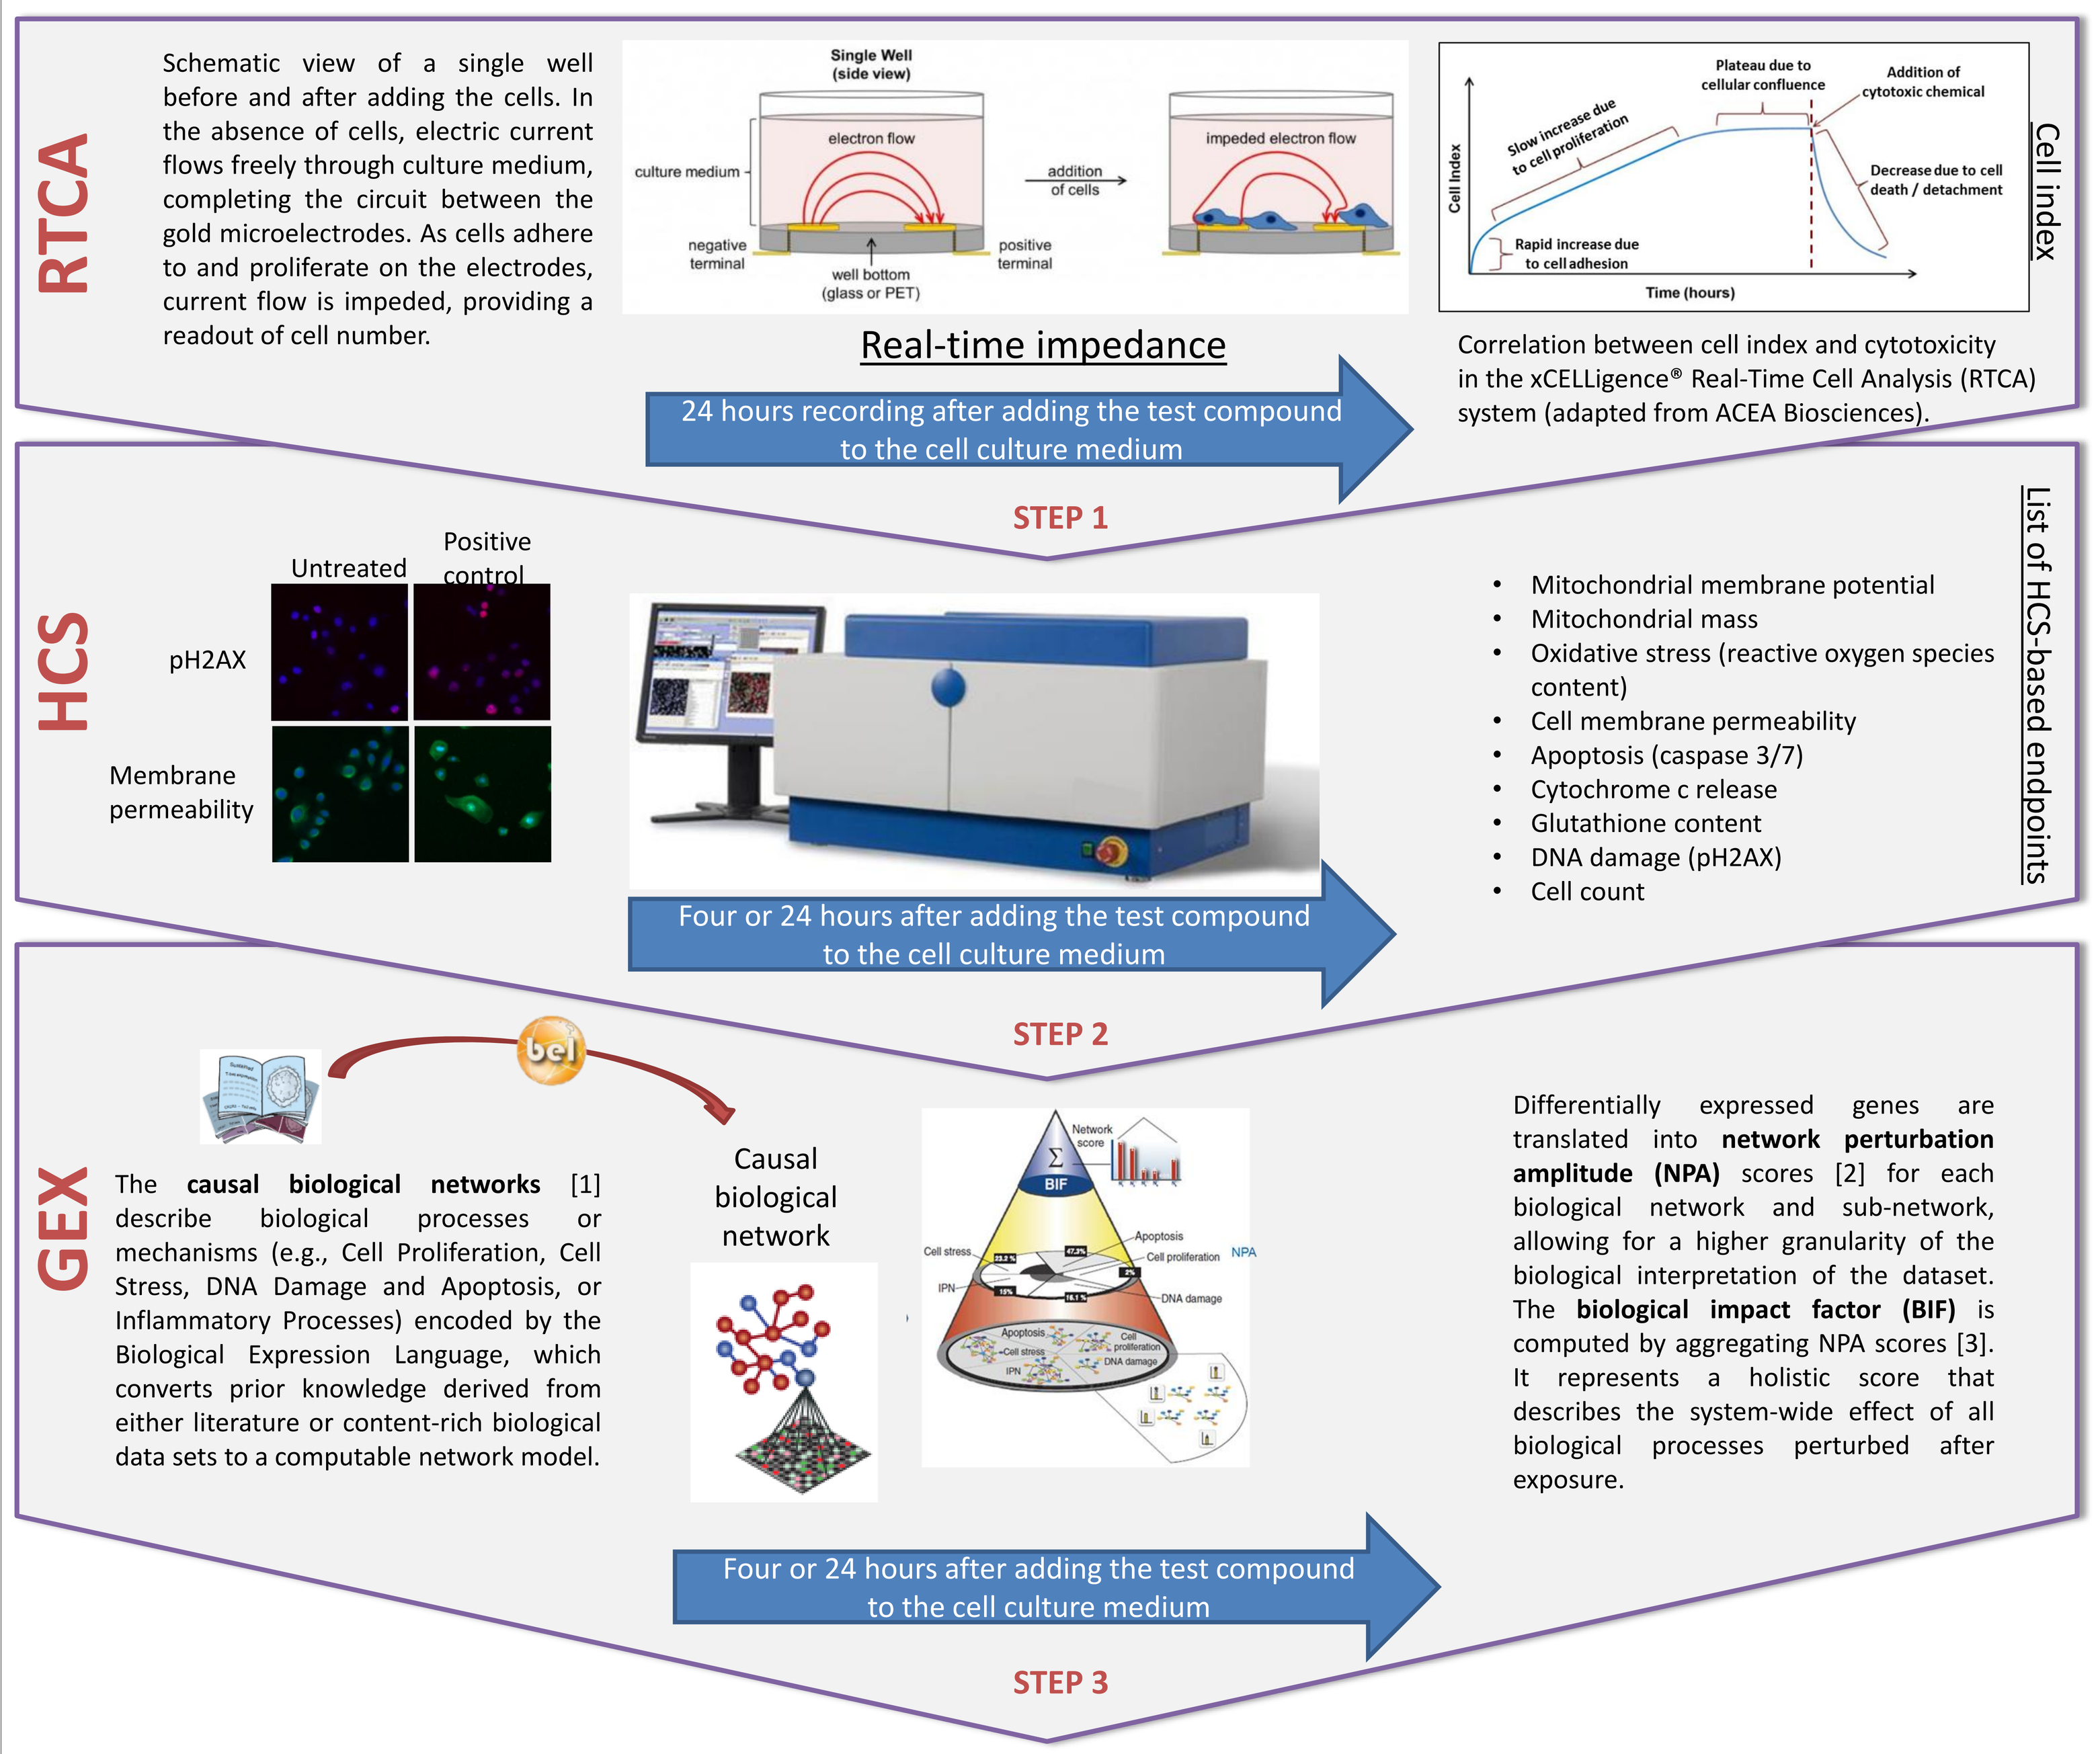
RTCA
Schematic view of a single well before and after adding the cells. In the absence of cells, electric current flows freely through culture medium, completing the circuit between the gold microelectrodes. As cells adhere to and proliferate on the electrodes, current flow is impeded, providing a readout of cell number.
Single Well (side view)
culture medium
electron flow
addition of cells
impeded electron flow
negative terminal
well bottom (glass or PET)
positive terminal
Real-time impedance
24 hours recording after adding the test compound to the cell culture medium
Cell index
Slow increase due to cell proliferation
Plateau due to cellular confluence
Addition of cytotoxic chemical
Decrease due to cell death / detachment
Rapid increase due to cell adhesion
Time (hours)
Correlation between cell index and cytotoxicity in the xCELLigence® Real-Time Cell Analysis (RTCA) system (adapted from ACEA Biosciences).
STEP 1
HCS
Untreated
Positive control
pH2AX
Membrane permeability
Four or 24 hours after adding the test compound to the cell culture medium
List of HCS-based endpoints
Mitochondrial membrane potential
Mitochondrial mass
Oxidative stress (reactive oxygen species content)
Cell membrane permeability
Apoptosis (caspase 3/7)
Cytochrome c release
Glutathione content
DNA damage (pH2AX)
Cell count
STEP 2
GEX
The causal biological networks [1] describe biological processes or mechanisms (e.g., Cell Proliferation, Cell Stress, DNA Damage and Apoptosis, or Inflammatory Processes) encoded by the Biological Expression Language, which converts prior knowledge derived from either literature or content-rich biological data sets to a computable network model.
Causal biological network
BIF
Network score
Cell stress
IPN
Apoptosis
Cell proliferation
NPA
DNA damage
Differentially expressed genes are translated into network perturbation amplitude (NPA) scores [2] for each biological network and sub-network, allowing for a higher granularity of the biological interpretation of the dataset. The biological impact factor (BIF) is computed by aggregating NPA scores [3]. It represents a holistic score that describes the system-wide effect of all biological processes perturbed after exposure.
Four or 24 hours after adding the test compound to the cell culture medium
STEP 3

Figure 1. The Flavor Assessment Workflow is a three-step approach designed to assess the toxicity of flavor compounds in NHBE cells. STEP 1 corresponds to a dose-range finding experiment using a real-time, impedance-based measurement system that will determine the Tox Score (see Figure 4) of each flavor compound. STEP 2 provides further information on the mechanism of toxicity triggered by the flavor compound exposure and is based on high-content screening (HCS) image analysis (see Figures 5 and 6). Only compounds with a Tox Score lower than 1 were tested in STEP 2. Finally, STEP 3 complements the mechanistic understanding of the flavor exposure effect using a systems toxicology approach based on transcriptomic data and computable biological networks.

Results

RTCA

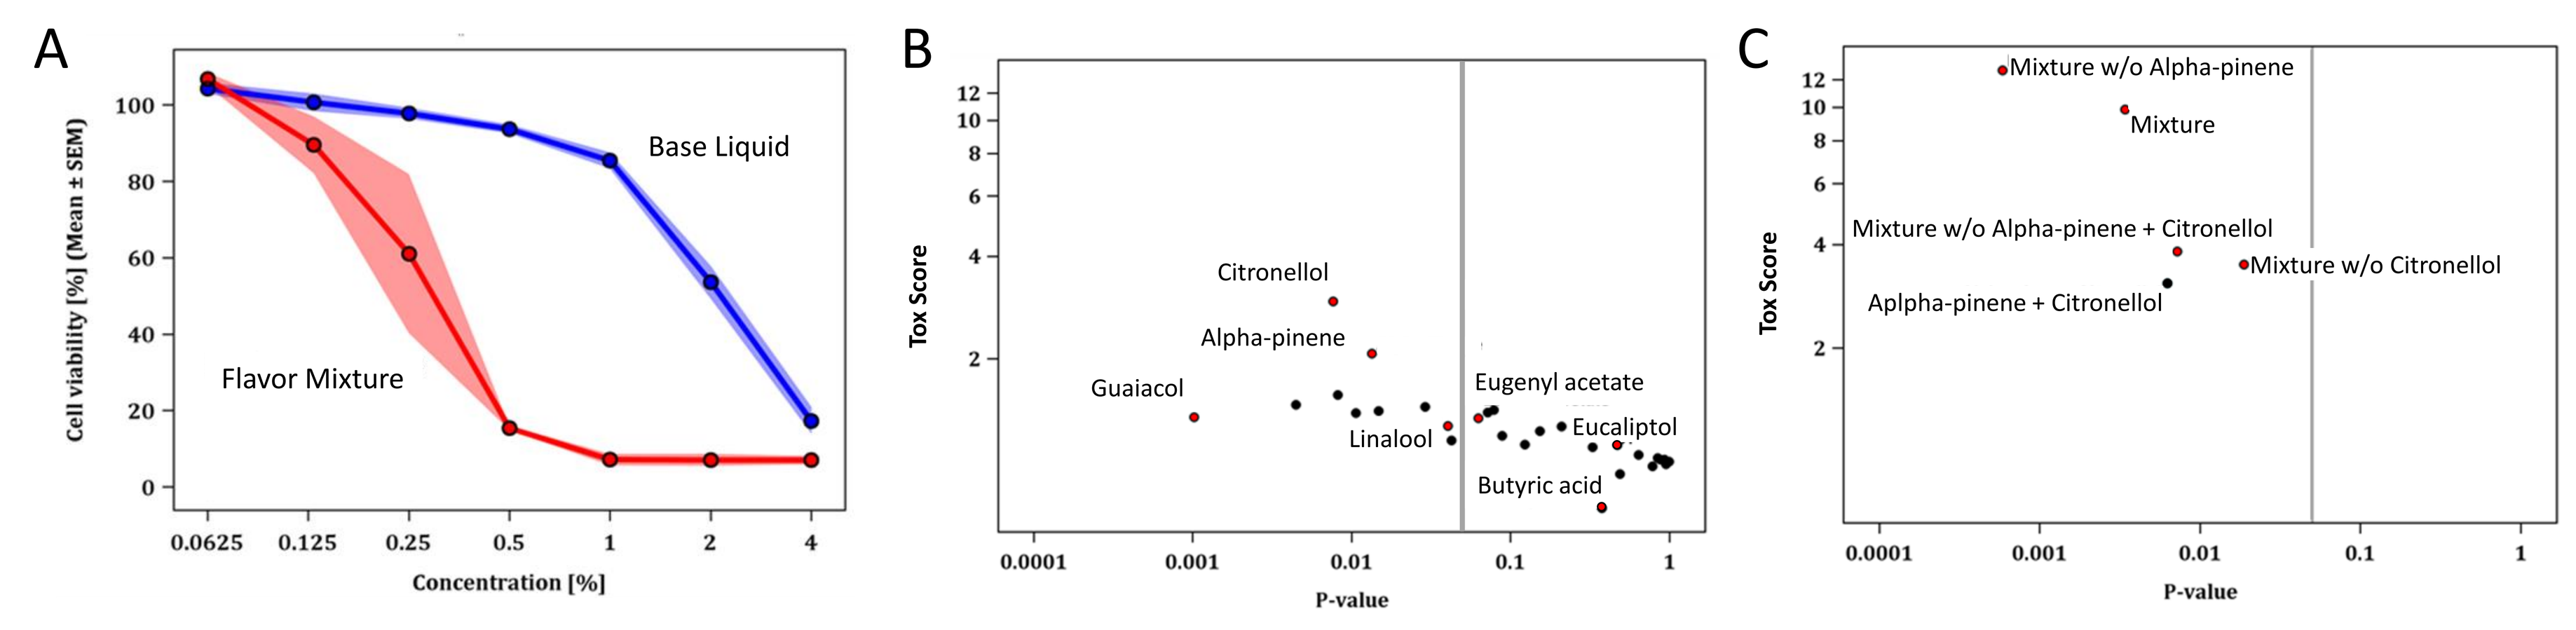
A
Cell viability [%] (Mean ± SEM)
Base Liquid
Flavor Mixture
Concentration [%]
B
Tox Score
Citronellol
Alpha-pinene
Guaiacol
Linalool
Eugenyl acetate
Eucaliptol
Butyric acid
P-value
C
Mixture w/o Alpha-pinene
Mixture
Mixture w/o Alpha-pinene + Citronellol
Mixture w/o Citronellol
Aplpha-pinene + Citronellol

Figure 2. (A) RTCA-based cell viability dose response curves for the 28-flavor mixture (in red) and the corresponding Base Solution (in blue). (B, C) Tox scores (*y*-axis) are represented as a function of their corresponding *p*-values computed (B) for each individual flavor present in the 28-flavor mixture and (C) for the flavor mixtures without either citronellol or alpha-pinene or both after a 24-hour exposure. The vertical line indicates a *p*-value of 0.05. Each dot corresponds to one flavor solution; those selected for HCS-based investigation are represented in red.

HCS

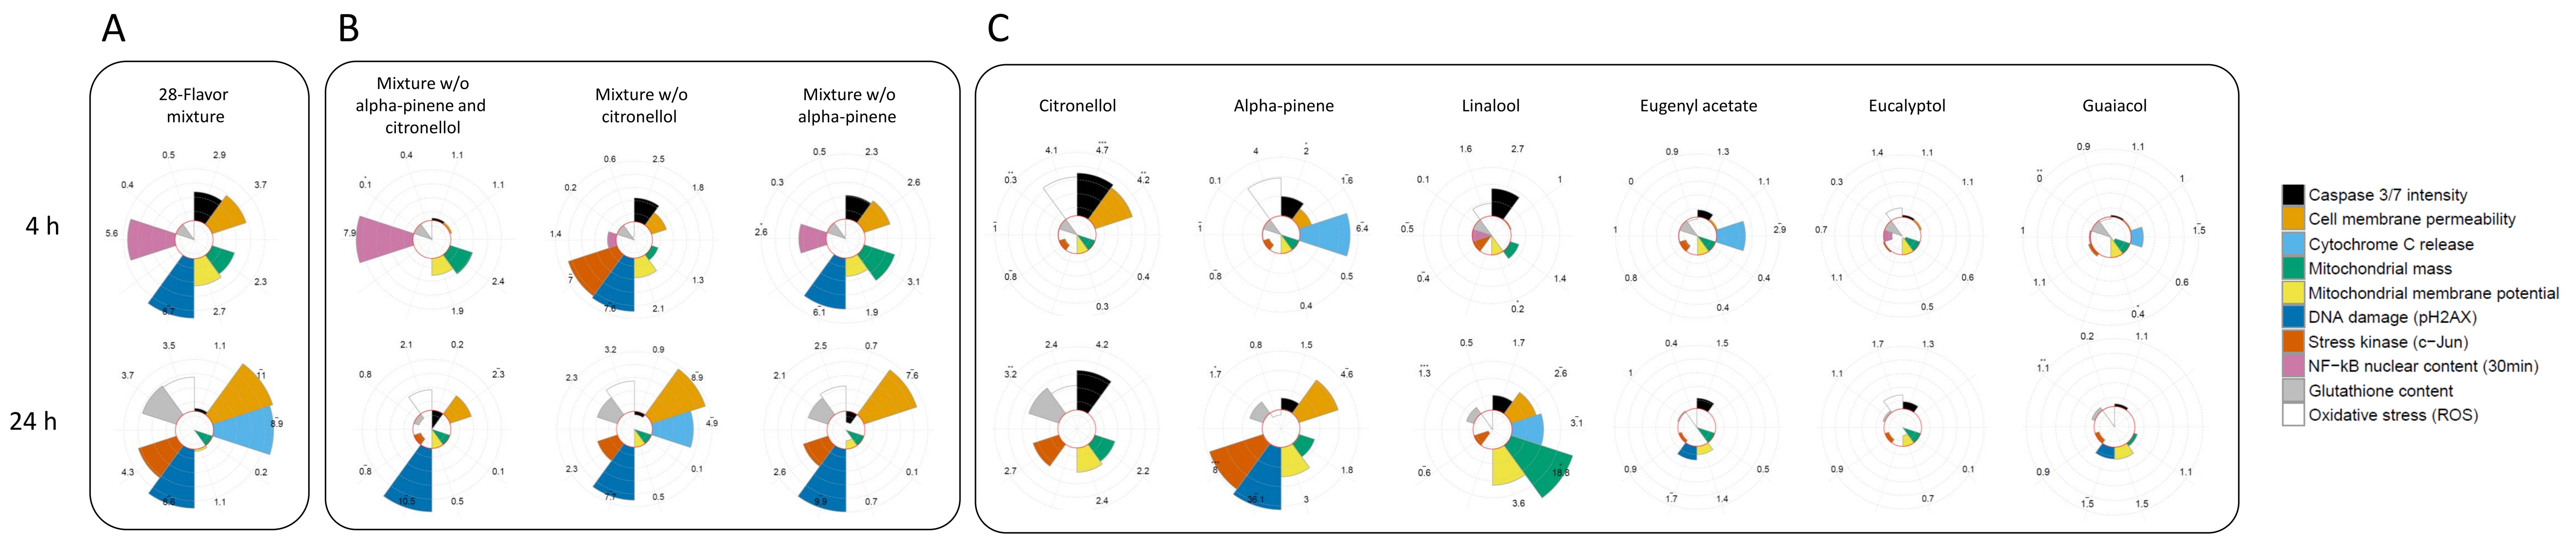
A
B
C
4 h
24 h
28-Flavor mixture
Mixture w/o alpha-pinene and citronellol
Mixture w/o citronellol
Mixture w/o alpha-pinene
Citronellol
Alpha-pinene
Linalool
Eugenyl acetate
Eucalyptol
Guaiacol
Caspase 3/7 intensity
Cell membrane permeability
Cytochrome C release
Mitochondrial mass
Mitochondrial membrane potential
DNA damage (pH2AX)
Stress kinase (c−Jun)
NF−kB nuclear content (30min)
Glutathione content
Oxidative stress (ROS)

Figure 3. Circular bar plots of HCS endpoint minimal effective concentration (MEC) ratios for (A) the 28-flavor mixture, (B) various individual flavors, and (C) flavor mixture without citronellol and/or alpha-pinene. Each MEC ratio (reported next to each segment of the circular chart for each HCS endpoint) was computed by dividing the mean Base Solution MEC (from n=3 replicates) by the mean flavor mix MEC. A *t*-test was computed with a null hypothesis: the Base Solution MEC mean is higher than the flavor mix MEC mean. The *t*-test *p*-values are reported as follows: *** < 0.001; ** < 0.01; * < 0.05. The “-“ sign on top of an MEC ratio denotes an imputed MEC value. Red circles correspond to an MEC of 1. Abbreviations: pH2AX, phosphorylated H2A histone family member X; NF-κB, nuclear factor kappa-light-chain-enhancer of activated B cells; ROS, reactive oxygen species.

GEX

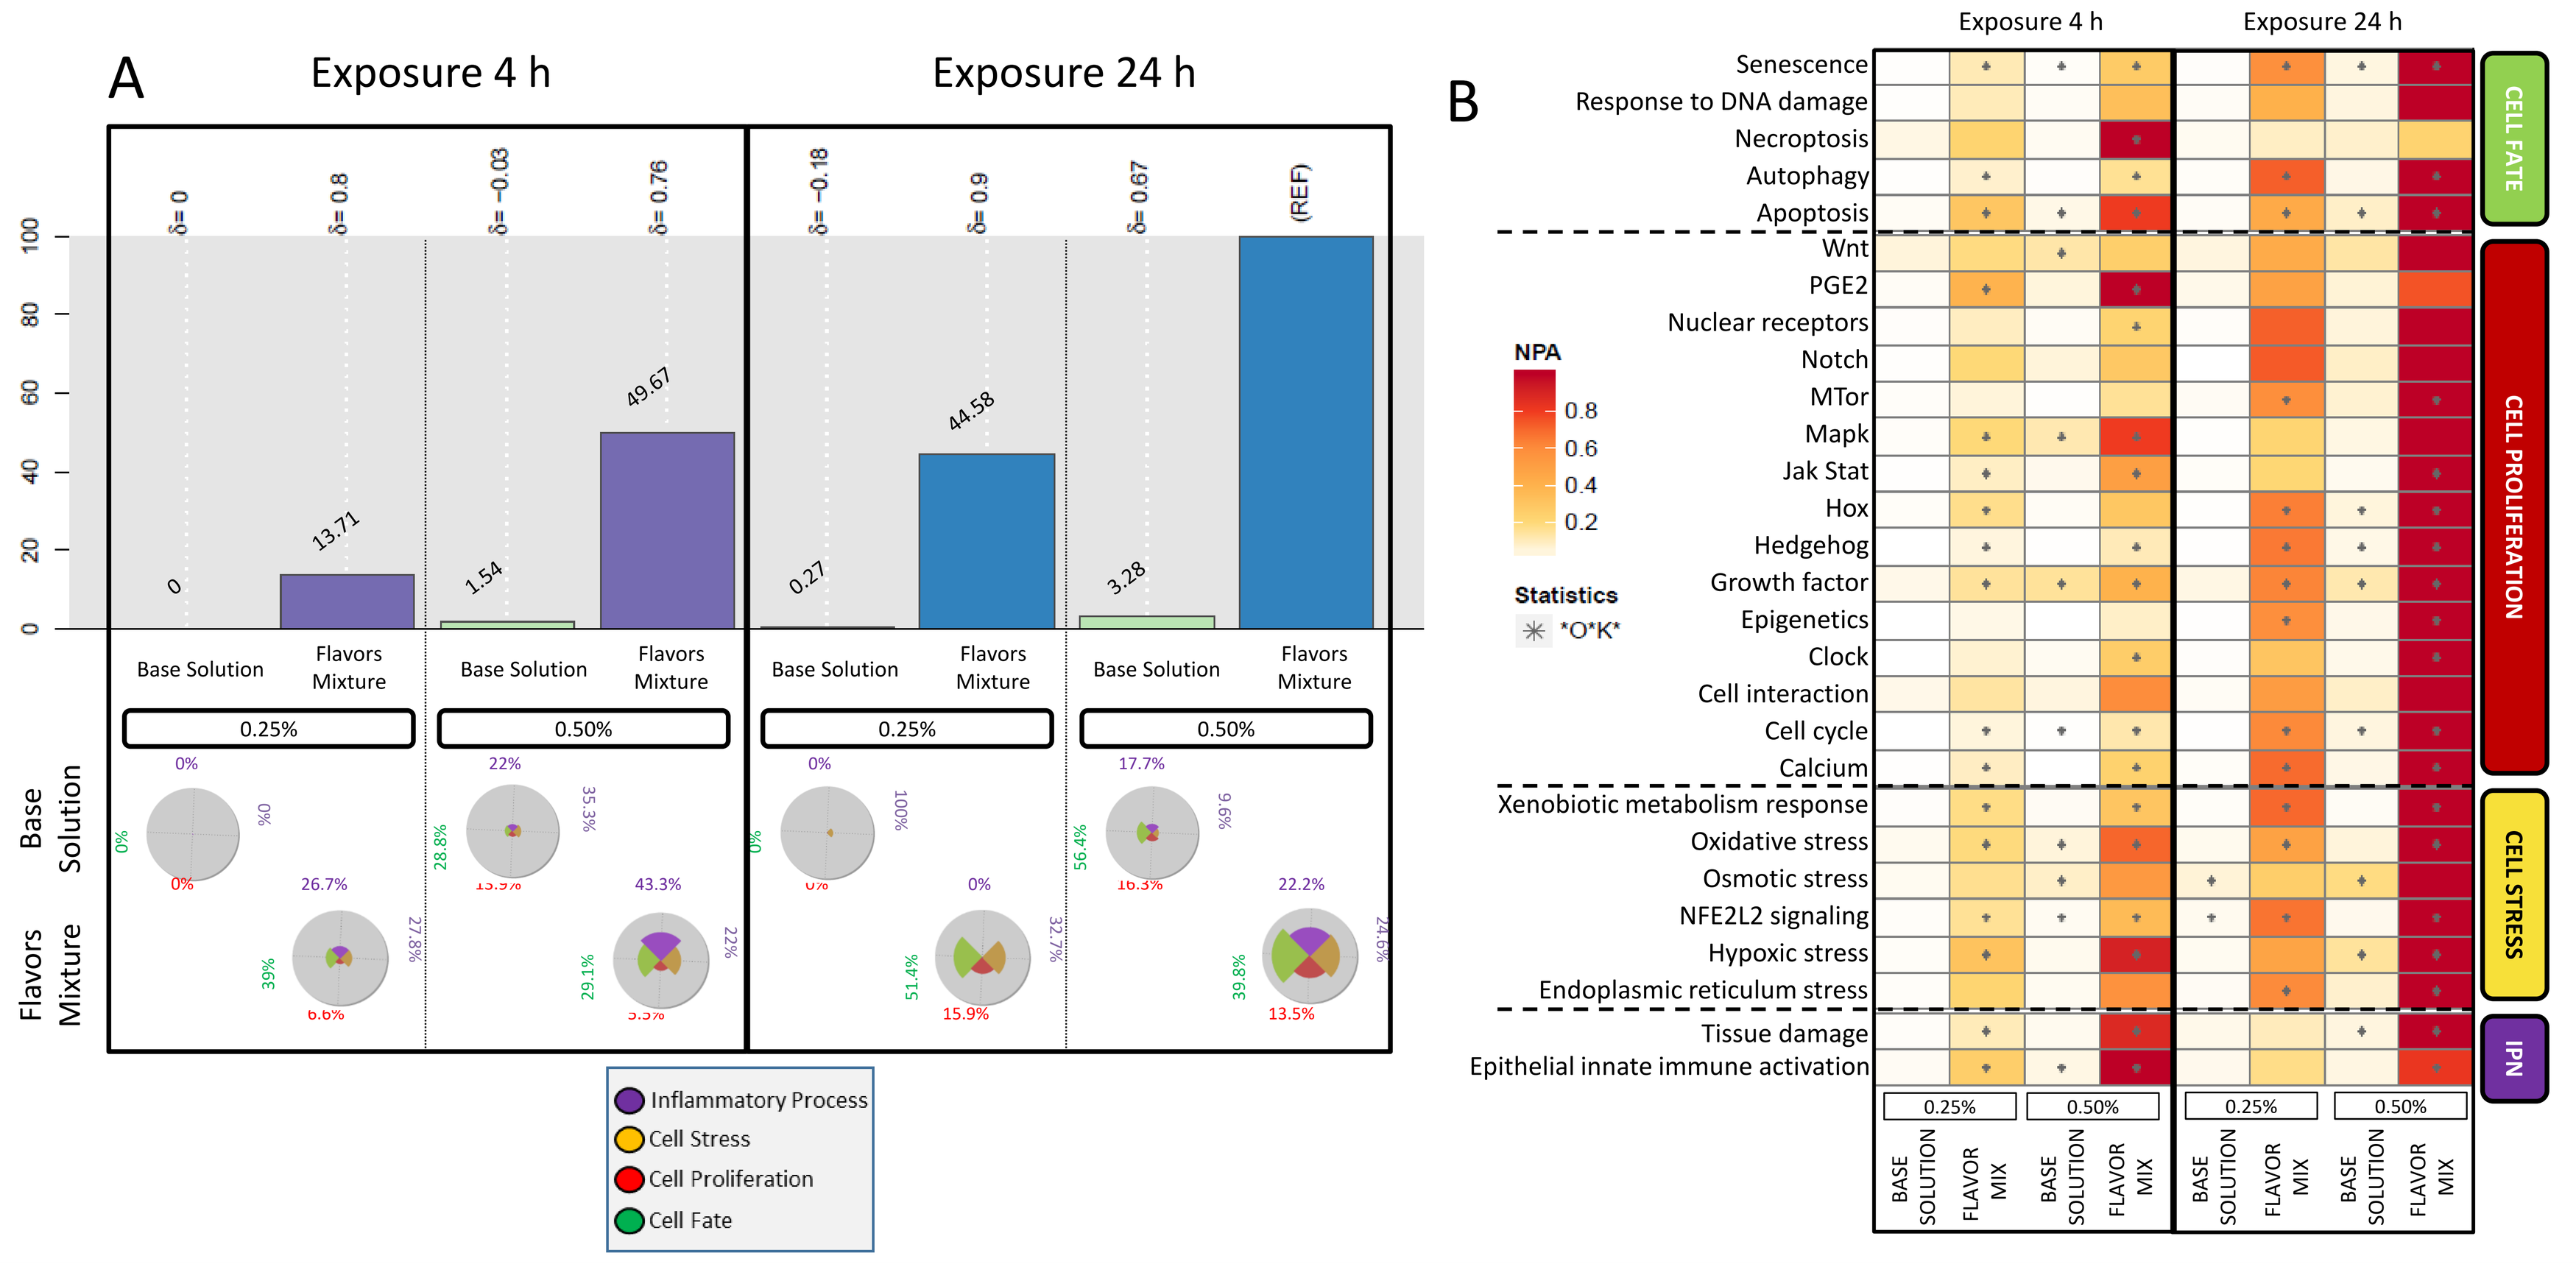
A
Exposure 4 h
Exposure 24 h
δ= 0
δ= 0.8
δ= −0.03
δ= 0.76
δ= −0.18
δ= 0.9
δ= 0.67
(REF)
%
0
13.71
1.54
49.67
0.27
44.58
3.28
Base Solution
Flavors Mixture
0.25%
0.50%
Base Solution
Flavors Mixture
Inflammatory Process
Cell Stress
Cell Proliferation
Cell Fate
B
Exposure 4 h
Exposure 24 h
Senescence
Response to DNA damage
Necroptosis
Autophagy
Apoptosis
Wnt
PGE2
Nuclear receptors
Notch
MTor
Mapk
Jak Stat
Hox
Hedgehog
Growth factor
Epigenetics
Clock
Cell interaction
Cell cycle
Calcium
Xenobiotic metabolism response
Oxidative stress
Osmotic stress
NFE2L2 signaling
Hypoxic stress
Endoplasmic reticulum stress
Tissue damage
Epithelial innate immune activation
NPA
Statistics
"O*K*"
CELL FATE
CELL PROLIFERATION
CELL STRESS
IPN
BASE SOLUTION
FLAVOR MIX

Figure 4. (A) Barplots (upper panel) represent relative BIF values (indicated above each bar) for NHBE cells exposed to the 28-flavor mixture or Base Solution using Cell Stress, Cell Proliferation, Cell Fate, and Inflammatory Processes networks. The percentages indicate the relative biological impact derived from the cumulated network perturbations caused by the treatment relative to the reference (defined as the treatment comparison showing the highest perturbation; REF = 100%). For each treatment comparison, the d value (–1 to 1) indicates how similar the underlying network perturbations are with respect to the reference. The pie charts (lower panel) represent the contributions of each network family. Percentage values in black indicate the dilution tested. (B) NPA heatmap of the subnetworks impacted by the Base Solution alone or by the flavor mixture at two dilutions (0.25% and 0.50% v/v) at the 4-hour and 24-hour time points. A network is considered perturbed if, in addition to the significance of the NPA score with respect to the experimental variation, the two companion statistics (O and K) that inform on the specificity of the NPA score with respect to the biology described in the network are also significant (as indicated by an asterisk). The darker the color, the stronger the perturbations. Abbreviation: IPN: Inflammatory Processes Network.

Table 2. Tox Scores determined using RTCA for each exposure condition tested.

$$\text{Tox Score} = \frac{\text{Base Solution EC50}}{\text{Flavor EC50}}$$

	Flavored solution	Tox Score	*p*-value
Individual	CITRONELLOL	2.948	0.007
	ALPHA-PINENE	2.076	0.013
	ALLYL HEXANOATE	1.568	0.008
	2-ACETYLTHIAZOLE	1.469	0.004
	METHYL ANTHRANILATE	1.448	0.029
	2-ETHYL-3,6-DIMETHYLPYRAZINE	1.417	0.078
	3-(METHYLTHIO) PROPIONALDEHYDE	1.409	0.014
	HEPTAN-2-ONE	1.394	0.071
	3-METHYL-2,4-NONANEDIONE	1.39	0.01
	GUAIACOL	1.352	0.001
	EUGENYL ACETATE	1.34	0.062
	LINALOOL	1.274	0.04
	4-(PARA-HYDROXYPHENYL)-BUTAN-2-ONE	1.269	0.209
	ETHYL MALTOL	1.228	0.152
	2-ACETYLPYRIDINE	1.191	0.088
	PHENETHYL ALCOHOL	1.156	0.042
	CYCLOTENE	1.122	0.123
	EUCALYPTOL	1.119	0.469
	FURANEOL	1.104	0.328
	ETHYL ACETATE	1.049	0.637
	CINNAMYL ALCOHOL	1.024	0.839
	MENTHONE	1.016	0.927
	GAMMA-VALEROLACTONE	1.014	0.871
	L-CARVONE	1.002	0.993
	METHYL SALICYLATE	0.982	0.948
	ETHYL FORMATE	0.971	0.777
	ISOBUTYL ALCOHOL	0.922	0.485
	BUTYRIC ACID	0.731	0.375
Mixture	FLAVOR MIXTURE w/o ALPHA-PINENE	12.8	0
	28-FLAVOR MIXTURE	9.85	0
	FLAVOR MIXTURE w/o CITRONELLOL and ALPHA-PINENE	3.82	0.01
	FLAVOR MIXTURE w/o CITRONELLOL	3.5	0.02
	CITRONELLOL and ALPHA-PINENE	3.09	0.01

Table 3. Phenotypic Scores and Endpoints count for the six individual flavors, 28-flavor mixture, and the three mixture variants that were evaluated using HCS endpoints. Corresponding Tox Score is also added.

$$\text{Phenotypic Score} = \frac{mean(\text{Base Solution MECs})}{mean(\text{Flavor MECs})}$$

Flavored solution	Phenotypic Score	Endpoints count
CITRONELLOL	3.16	12
28-FLAVOR MIXTURE	2.54	18
ALPHA-PINENE	2.52	15
FLAVOR MIXTURE w/o ALPHA-PINENE	1.75	16
FLAVOR MIXTURE w/o CITRONELLOL	1.44	19
LINALOOL	1.30	13
FLAVOR MIXTURE w/o CITRONELLOL and ALPHA-PINENE	1.14	14
EUCALYPTOL	1.09	14
GUAIACOL	0.84	15
EUGENYL ACETATE	0.79	16

Conclusions

- The approach described in this poster is based on three pillars: (i) RTCA, (ii) a panel of phenotypic HCS endpoints, and (iii) a gene expression analysis. For each pillar, computationally derived scores were developed and used to quantify (i) the toxicity (Tox Score), (ii) the phenotypic impact (Phenotypic Score) and (iii) the transcriptomic/mechanistic effect (BIF) of an exposure.
- The 28-flavor mixture appeared to induce higher cytotoxicity than the corresponding flavorless base solution. By individually testing each of the flavors, citronellol and alpha-pinene appeared to be the main contributors to the overall mixture cytotoxicity.
- When individually removed from the mixture, cytotoxic contribution was confirmed only for citronellol, while alpha-pinene removal did not lead to any change of mixture cytotoxicity.
- The remaining flavors, which showed limited cytotoxicity when tested individually, appeared to significantly contribute to the overall mixture cytotoxicity (synergistic effect).
- Using an artificial mixture of 28 flavors dissolved in a base solution, we showed that this method is suitable to identify candidate constituents that are accountable for the toxicity of a mixture (such as citronellol in our case study).

To know more about the HCS data analysis pipeline, visit poster number: P101

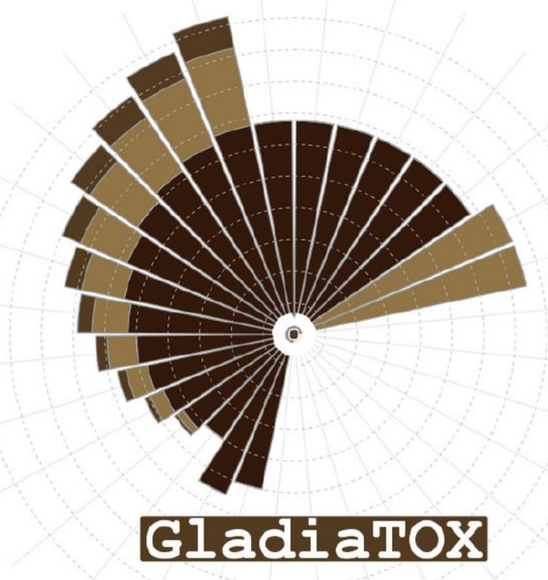
GladiaTOX

Data will be soon available on: www.intervals.science

INTERVALS
ADVANCING SCIENCE FOR A SMOKE-FREE WORLD

SOT 2019 I Baltimore I March 10-14 2019

Competing Financial Interest – The research described in this poster was sponsored by Philip Morris International