Development and testing of the Independent Holistic Air-Liquid Aerosol Exposure System (InHALES)

S. Steiner, P. Hervé, S. Majeed, C. Pak, A. Kuczaj, J. Hoeng

PMI R&D, Philip Morris Products S.A., Quai Jeanrenaud 5, CH-2000 Neuchatel, Switzerland

Background

- The delivery kinetics of volatile and particulate aerosol constituents as well as of particles of different sizes vary between different regions of the human respiratory tract *[1, 2, 3]*.
- Inhaled aerosols evolve in terms of particle size distribution and chemical composition when passing from the site of their generation through the respiratory tract to the alveolar spaces and back *[1, 2, 3]*.
- Available *in vitro* aerosol exposure systems are not able to capture this complexity *[4, 5]*. The aerosol fractions they deliver to cell cultures are therefore not, or only partly, representative for the *in vivo* situation.
- This may decrease the relevance of *in vitro* aerosol exposure experiments, especially when using complex cell cultures that are able to respond to physical and chemical stimuli in a highly differentiated manner.
- We developed an *in vitro* aerosol exposure system that mimicks structural and functional aspects of the human respiratory tract: the *In*dependent *H*olistic *A*ir *L*iquid *E*xposure *S*ystem (InHALES) *[6]*.
 - Independent: it is capable of actively inhaling aerosols, smoking cigarettes, or operating inhalers without the need for special aerosol generators or diluters.
 - Holistic: it consists of modules representing the relevant regions of the respiratory tract; the existing prototype consists of oral cavity, laryngopahrynx, trachea, main bronchi, bronchioles, and lung lumen modules.
 - Air-liquid exposure system: it was specifically designed for aerosol exposures at the air-liquid interface.
- A prototype of the system has recently been built (**Figure 1**), and initial system characterization, including exposures of three-dimensional organotypic models of the human airway epithelia, was performed.

The InHALES system

Mouth pump (Figure 1.1).

- 0–140 mL; maximum flexibility for puff generation.
- Connection to the tracheal model sealed by a butterfly valve.
- Five slots for placing cell cultures.

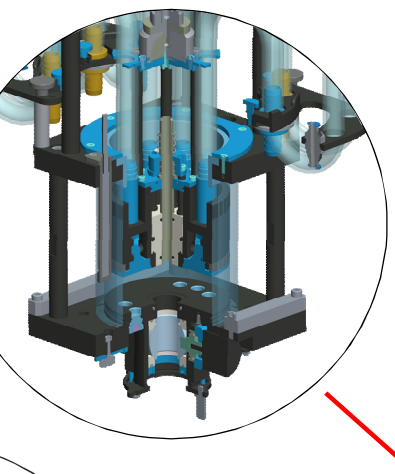

Figure 1.1: The mouth pump in opened position.

Lung pumps (Figure 1.2)

- 0–6000 mL; full flexibility with respect to timing, profile, and residual volume.
- 5 slots for cell cultures per pump.

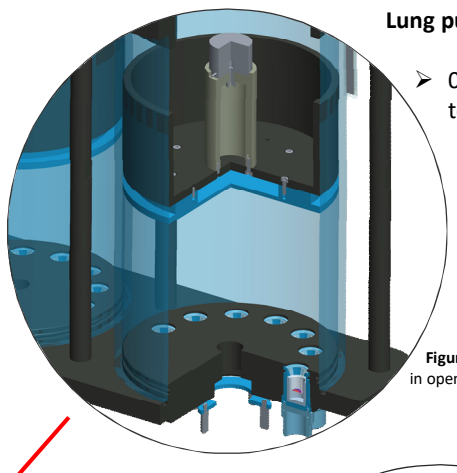

Figure 1.2: The lung pumps in opened position.

Trachea and main bronchi (Figure 1.3)

- Size and geometry based on human physiology.
- Cell cultures on hydrogels, maximizing flexibility of positioning and minimizing culture size.

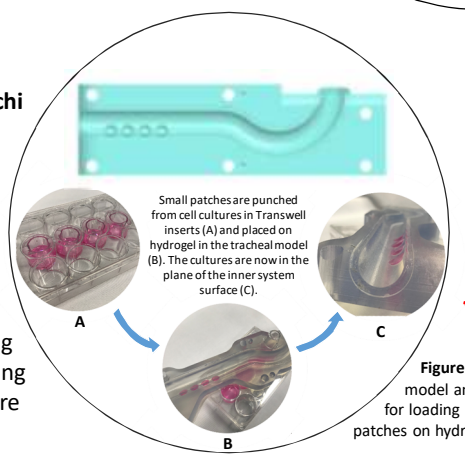
Small patches are punched from cell cultures in Transwell inserts (A) and placed on hydrogel in the tracheal model (B). The cultures are now in the plane of the inner system surface (C).
A
B
C

Figure 1.3: The tracheal model and the procedure for loading cell culture patches on hydrogels.

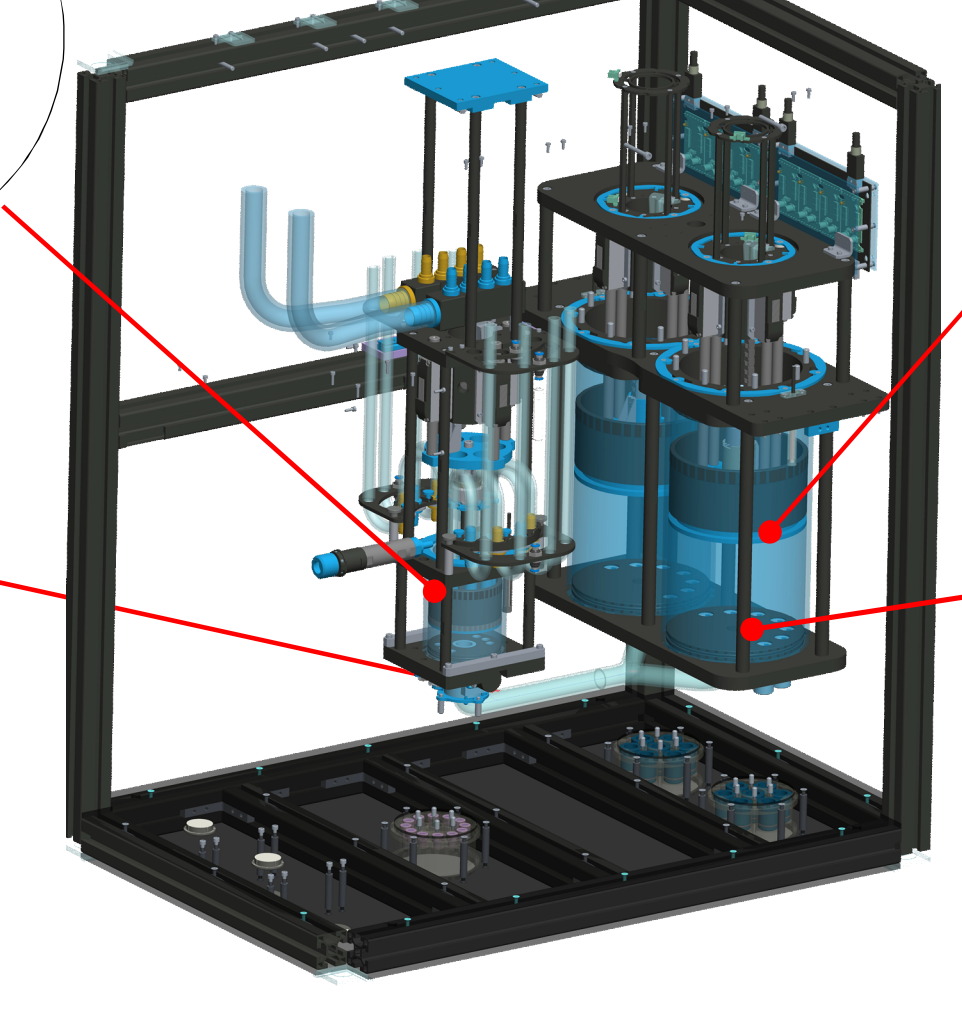

Figure 1: Total view of the InHALES system. The system is modular; individual parts (mouth pump, trachea, main bronchi, bronchial tree, lung pumps) can be modified or exchanged without changing the overall system structure or function.

Bronchial tree (Figure 1.4)

- Plugged inside the lung pumps (not included in Figure 1).
- 5 generations of bifurcations realized in the prototype.
- Key geometrical parameters based on Weibel's model *[8].*

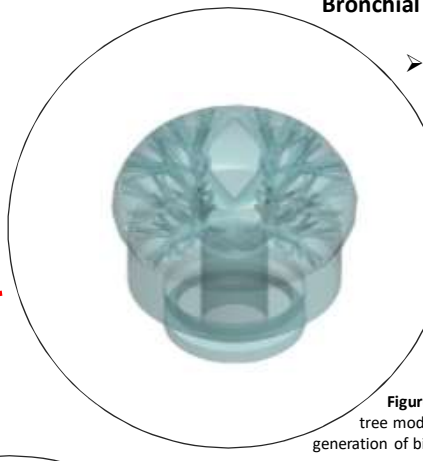

Figure 1.4: The bronchial tree model reaching the 5th generation of bifurcation.

System operation (Figure 1.5)

- Orchestrated operation of valves and pumps.
- Simulation of virtually any breathing pattern is possible.

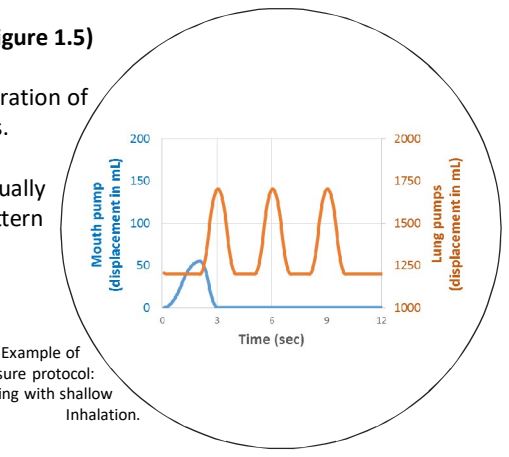
Mouth pump (displacement in mL)
Lung pumps (displacement in mL)
Time (sec)
200
150
100
50
0
2000
1750
1500
1250
1000
0
3
6
9
12

Figure 1.5: Example of an exposure protocol: smoking with shallow Inhalation.

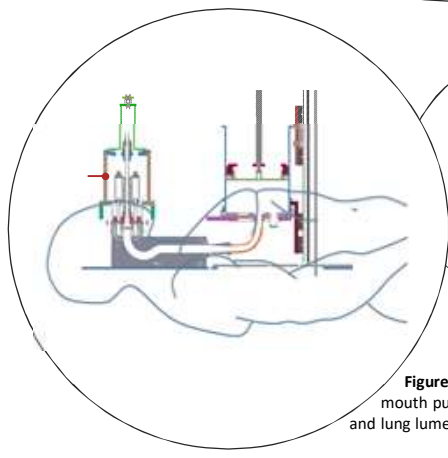

Figure 1.6: The arrangement of mouth pump, trachea, main bronchi, and lung lumen mimics human physiology.

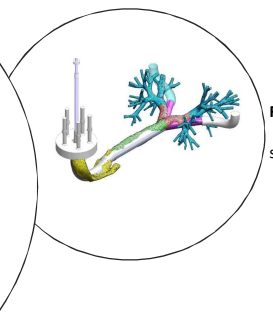

Figure 1.7: Trachea and main bronchi are based on human physiology in size and geometry:
- Brno lung cast model *[7]*
- Weibel's model *[8]*

Test exposures, results, and discussion

Test exposure settings: The system "puffed" and "inhaled" a fluorescent test aerosol (a nebulized mixture of propylene glycol [PG], glycerol [G], and water, labelled with disodium fluorescein [DSF]) or smoke generated from 3R4F cigarettes (University of Kentucky). 4–5 repetitions were conducted; in each repetition, 60 puffs were delivered.

Two different exposure protocols were applied:

- **Shallow inhalation** (see Figure 1.5): 1200 mL residual volume in lung pumps, 2 seconds puff generation, immediate puff inhalation during 1 second along with 500 mL clean air, 2 empty inhalations (no puff) within 6 seconds (Figure 1.5).
- **Deep inhalation** (Figure 2): 1200m mL residual volume in lung pumps, 2 seconds puff generation, immediate puff inhalation during 1 second along with 4,600 mL clean air.

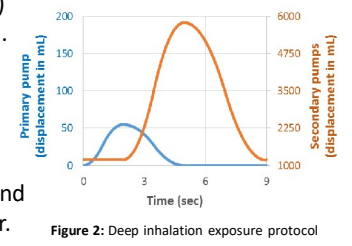
Primary pump (displacement in mL)
Secondary pumps (displacement in mL)
Time (sec)
200
150
100
50
0
6000
4750
3500
2250
1000
0
3
6
9

Figure 2: Deep inhalation exposure protocol

During exposures, the complete system was heated to 37°C, and inhaled air was brought to a relative humidity of 95%.

Aerosol delivery was investigated by exposing samples of 300 µL phosphate-buffered saline (PBS), followed by quantification of nicotine and 8 representative carbonyl compounds by liquid chromatography–mass spectrometry (for 3R4F smoke) or DSF (for PG/G aerosol) by fluorometry. PBS was exposed in cell culture inserts in the pumps only (not in the trachea).

Cell culture exposures were conducted along with PBS exposures. EpiOral cultures (MatTek, Ashland, USA) were exposed in the mouth pump, patches of MucilAir cultures (Epithelix SarL, Geneva, Switzerland) in the trachea, and A549 cultures in the lung pumps.

Biological endpoint assessment: 24 hours after exposures, cytotoxic effects (quantification of extracellular lactate dehydrogenase [LDH], Cytotoxicity Detection KitPLUS [Roche, Basel, Switzerland]) and cell culture morphology (hypoxanthine and eosine [H&E] staining for EpiOral and MucilAir, Immunofluorescence for A549) were assessed.

Particle size distributions in the aerosols were measured at different locations in the system using a TSI 3321 aerodynamic particle sizer.

Aerosol delivery and evolution

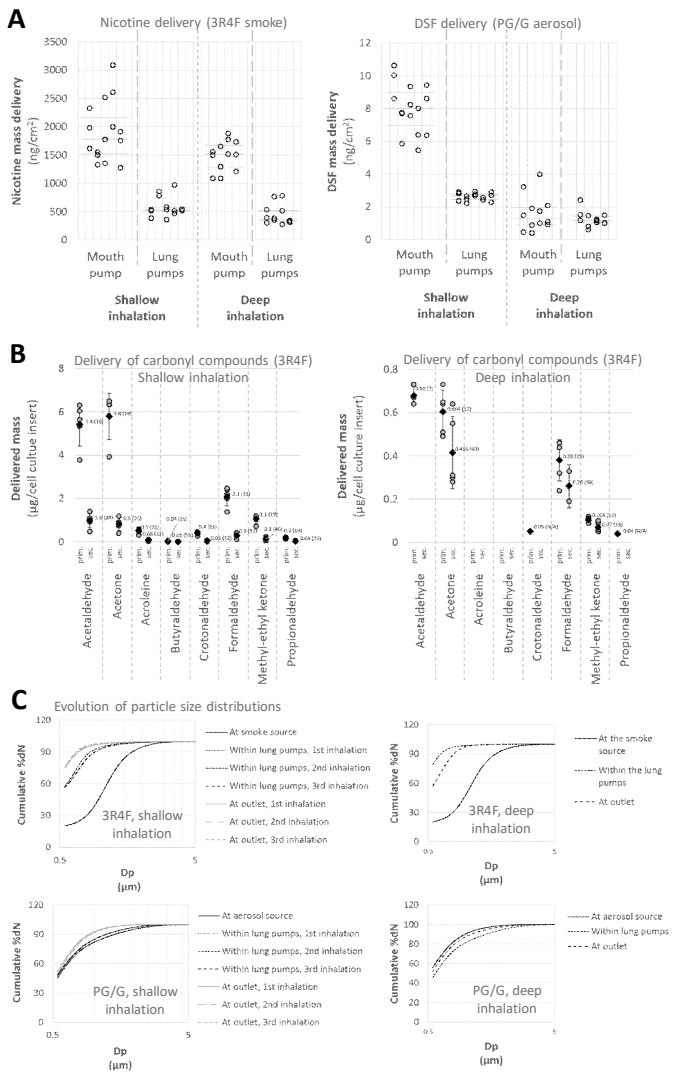
A
Nicotine delivery (3R4F smoke)
Nicotine mass delivery (ng/cm²)
DSF delivery (PG/G aerosol)
DSF mass delivery (ng/cm²)
Mouth pump
Lung pumps
Shallow inhalation
Deep inhalation
B
Delivery of carbonyl compounds (3R4F) Shallow inhalation
Delivery of carbonyl compounds (3R4F) Deep inhalation
Delivered mass (µg/cell culture insert)
Acetaldehyde
Acetone
Acroleine
Butyraldehyde
Crotonaldehyde
Formaldehyde
Methyl-ethyl ketone
Propionaldehyde
C
Evolution of particle size distributions
Cumulative %dN
Dp (µm)
3R4F, shallow inhalation
3R4F, deep inhalation
PG/G, shallow inhalation
PG/G, deep inhalation
At smoke source
Within lung pumps, 1st inhalation
Within lung pumps, 2nd inhalation
Within lung pumps, 3rd inhalation
At outlet, 1st inhalation
At outlet, 2nd inhalation
At outlet, 3rd inhalation
At aerosol source
Within the lung pumps
At outlet

Figure 3: A) delivery of nicotine (3R4F exposures) and DSF (PG/G exposures) within the mouth and lung pumps (4–5 repetitions each with 3 samples exposed). **B)** according data on carbonyl deposition during 3R4F exposures. Error bars show standard deviations. **C)** Evolution of (cumulative) particle size distributions during the smoke's/aerosol's passage through the system.

Biological endpoints

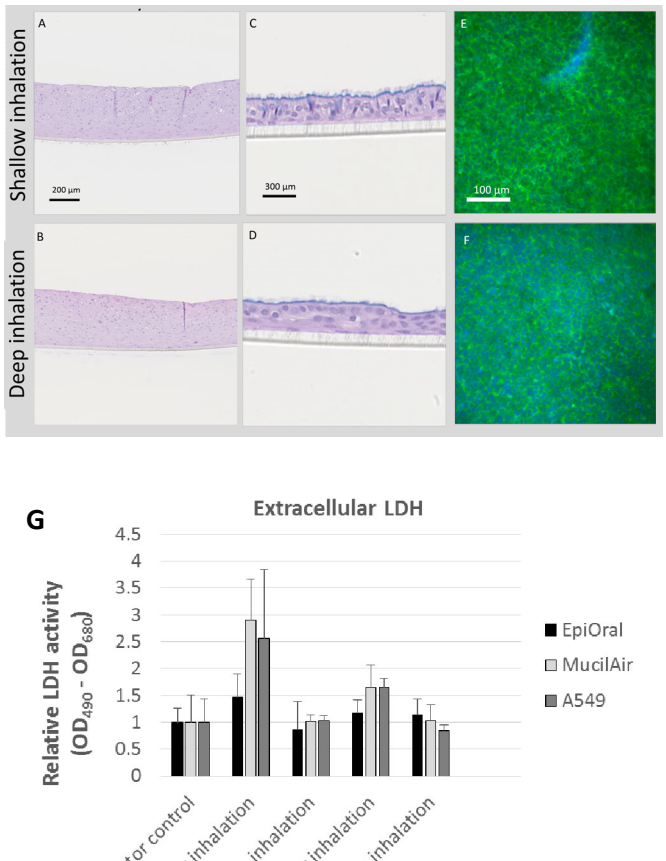
Shallow inhalation
Deep inhalation
A
B
C
D
E
F
200 µm
300 µm
100 µm
G
Extracellular LDH
Relative LDH activity (OD_{490} - OD_{680})
4.5
4
3.5
3
2.5
2
1.5
1
0.5
0
EpiOral
MucilAir
A549
Incubator control
3R4F, shallow inhalation
3R4F, deep inhalation
PG/G, shallow inhalation
PG/G, deep inhalation

Figure 4: Biological responses to the test exposures. **A)** and **B)** H&E-stained EpiOral tissues exposed under shallow inhalation or deep inhalation settings, respectively. **C)** and **D)** H&E-stained MucilAir tissue patches. **E)** and **F)** A549 cultures stained for nucleic acids (hoechst, blue) and F-actin (Phalloidine, green). PG/G-exposed tissues were selected as example. **G)** Extracellular LDH detected in the basolateral culture medium or the hydrogel, expressed relative to the incubator controls. In Triton-X-100-treated cultures, values of 230 (EpiOral), 30 (MucilAir) and 61 (A540) were measured. Error bars represent standard deviations.

- The system's applicability for controlled aerosol delivery, in particular in combination with biological test systems, was demonstrated
 - No system-related adverse effects of exposures were detected, and cell cultures were responsive to harmful stimuli (3R4F smoke).
 - Changes in system settings or test aerosols translate into differential biological responses.
- Aerosol delivery within the system is stable and repeatable within the expected range. The geometry of inlets at the pumps will be optimized, however, which we expect to increase the delivery uniformity and repeatability.
- We observed complex aerosol dynamics in the system, which is considered the result of particle sizes, the partitioning of aerosol constituents between the particulate and the gas phase, and the complexity of the system.
 - System complexity translates into complex aerosol dynamics and delivery.
 - A 1:1 comparison to the aerosol delivery within the human respiratory tract was not yet included, as relevant parameters are not covered in the exisiting prototype system (limited complexity of the bronchial tree model and lung surface area).
- System geometry will be further optimized. In particular, the complexity of the airway tree and the inner surface area of the lung pump will be increased.

References:

1. Carvalho TC, Peters JI, Williams RO. *International Journal of Pharmaceutics* 2011, **406**(1–2):1-10.
2. Heyder J. *Proceedings of the American Thoracic Society* 2004, **1**(4):315-320.
3. Pankow JF. *Chem Res Toxicol* 2001, **14**(11):1465-1481.
4. Thorne D, Adamson J. *Exp Toxicol Pathol* 2013, **65**(7):1183-1193.
5. Paur HR, Mulhopt S, Weiss C, Diabate S. *J Verbrauch Lebensm* 2008, **3**(3):319-329.
6. Hervé, P. In vitro Experiments: Development of a New Aerosol Exposure System. Master Thesis at the école d'ingénieurs CESI, Saint-Nazaire, 2017
7. Ochs M, Weibel ER (eds.). McGraw-Hill; 2008.
8. Nordlund M, Belka M, Kuczaj AK, Lizal F, Jedelsky J, Elcner J, Jicha M, Sauser Y, Le Bouhellec S, Cosandey S. *Inhalation toxicology* 2017, **29**(3):113-125.

PMI SCIENCE
PHILIP MORRIS INTERNATIONAL

The research described in this poster was sponsored by Philip Morris International