Establishing the baseline effects of non-flavoured e-liquids and flavour compounds in the *in vitro* micronucleus assay

DJ Smart, FR Helbling, E Nallet, D McHugh, and P Vanscheeuwijck

PMI R&D, Philip Morris Products S.A., Quai Jeanrenaud 5, CH-2000 Neuchâtel, Switzerland

Introduction and Goal

- A two-part screening study was undertaken with the goal of establishing the baseline effects of 1) non-flavoured e-liquids (NLs) and 2) a subset of flavour compounds in the flow cytometry version of the *in vitro* micronucleus (MNvit) assay. In addition, a ToxTracker® analysis on the same flavours was carried out in order to benchmark its performance against the MNvit assay.

- Part 1: 20 mg/ml nicotine-containing NLs were composed of the following ratios (%) of propylene glycol (PG) and vegetable glycerin (VG): NL-A (70PG:20VG), NL-B (40PG:40VG), NL-C (20PG:73VG), NL-D (100PG), and NL-E (100VG). A nicotine-free NL similar in composition to NL-A was also included: NL-F (70PG:20VG).

- Part 2: 11 flavour compounds were selected based on their mammalian genotoxic potential *in vitro*,

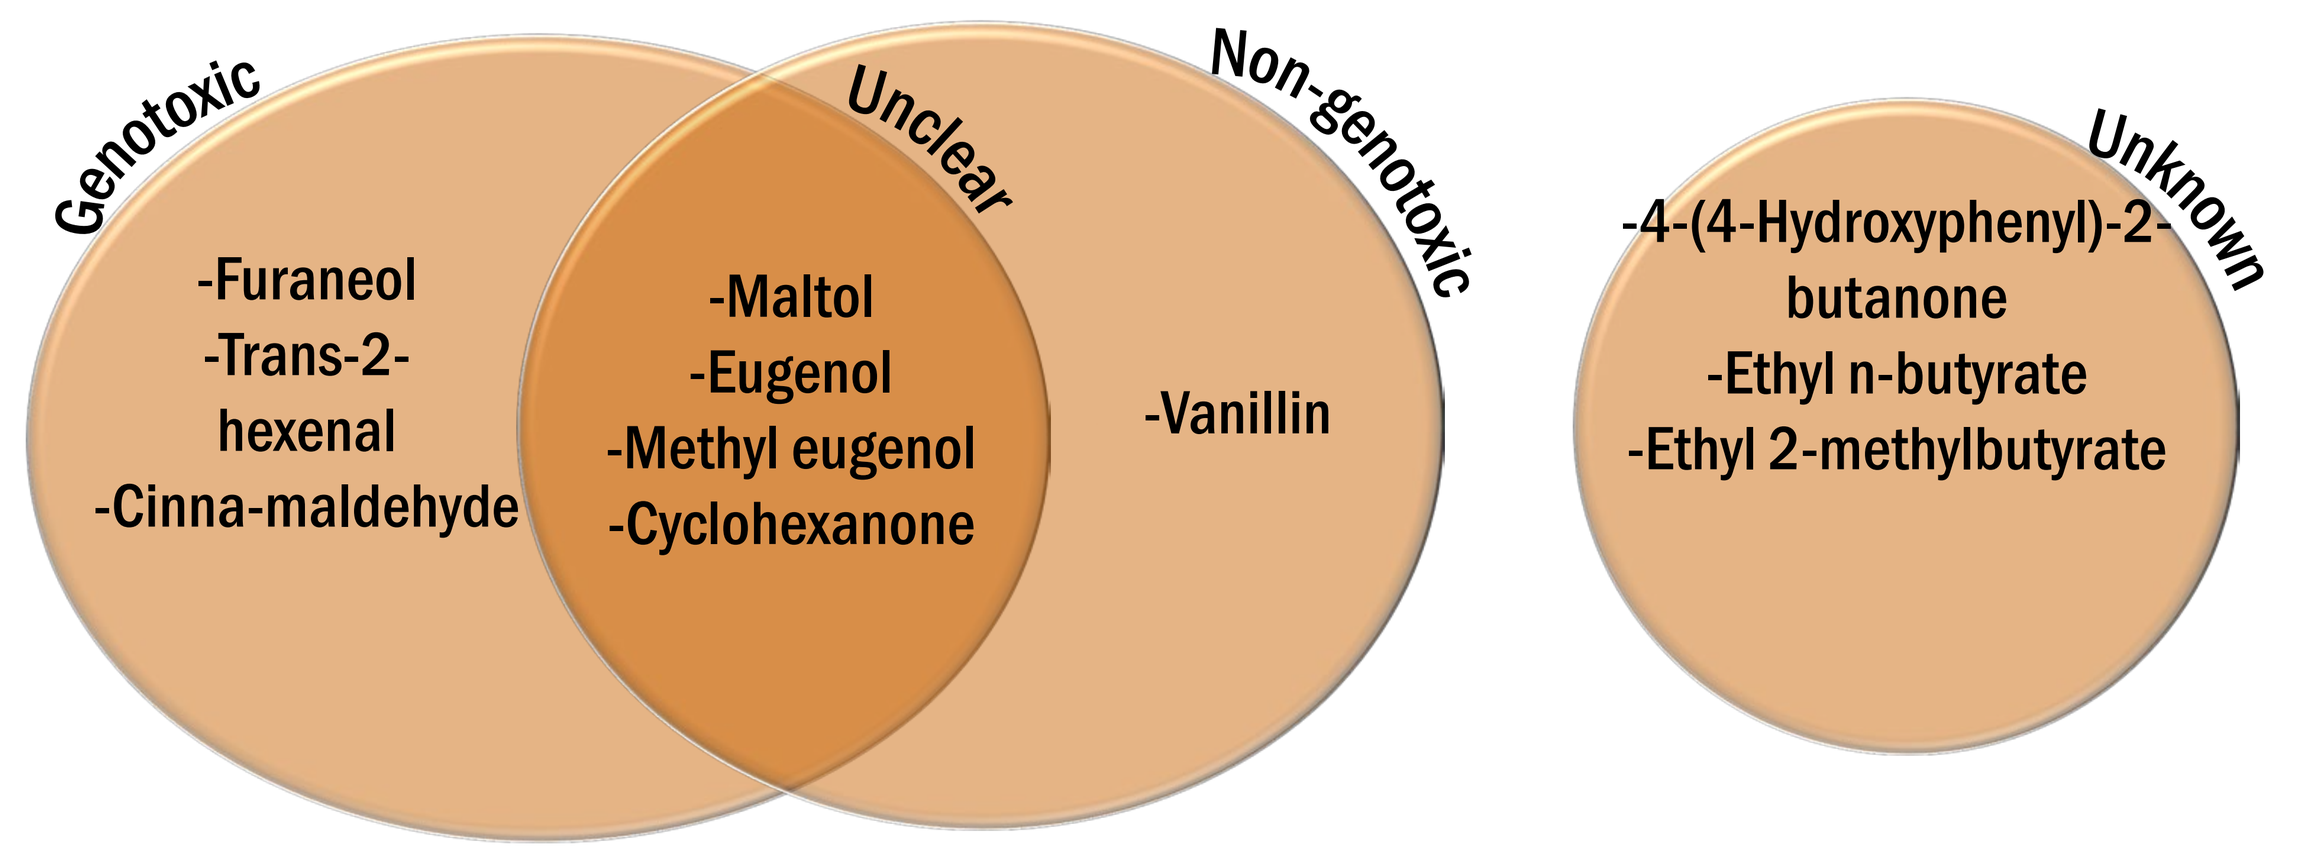
Genotoxic
Unclear
Non-genotoxic
Unknown
-Furaneol
-Trans-2-hexenal
-Cinna-maldehyde
-Maltol
-Eugenol
-Methyl eugenol
-Cyclohexanone
-Vanillin
-4-(4-Hydroxyphenyl)-2-butanone
-Ethyl n-butyrate
-Ethyl 2-methylbutyrate

Materials and Methods

- **Cell line:** Chinese hamster ovary-Wolff Bloom Litton cell line (CHO-WBL; Merck Research Laboratories, USA). Cells were seeded in 96-well plates (Nunclon®, USA) at 4,500 cells/well for 24 hours (h) prior to treatment in McCoy's 5A + GlutaMAX™ medium + 3% (+S9) or 10% (24 h) HI-FBS + 100 units/ml Penicillin + 0.1 mg/ml Streptomycin and incubated at 37°C with 5% CO_2 and 70% relative humidity.

- **Test substances:** NLs were prepared in-house using the highest grade PG, VG, H_2O and (-)-nicotine chemicals. Flavour compounds were also of the highest grade possible and solubilised in DMSO prior to treatment.

- **Extreme culture conditions assessment:** Changes in the pH and osmolality of the cell culture medium immediately post-exposure to NLs were measured using a pH meter and osmometer, respectively.

- **MNvit treatment conditions:** NLs were only evaluated in the 24 h treatment condition and up to the cytotoxicity limit of the assay, while flavours were evaluated in both 4 h +S9 and 24 h treatment conditions using a general limit concentration of 1 mM. S9: Aroclor-1254-induced male Sprague Dawley rat liver (Moltox, USA).

- **MNvit testing procedure (modified from OECD TG 487, 2016):**

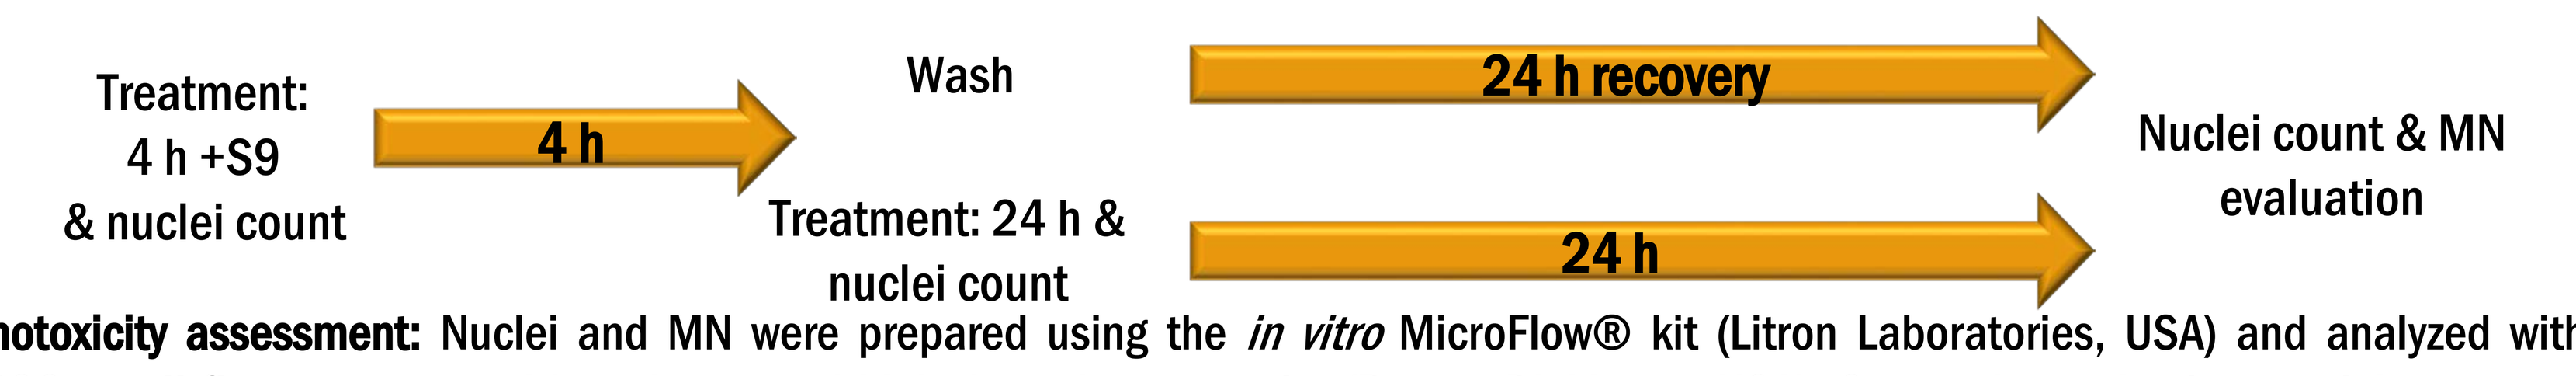
Treatment:
4 h +S9
& nuclei count
4 h
Wash
Treatment: 24 h &
nuclei count
24 h recovery
24 h
Nuclei count & MN
evaluation

- **Genotoxicity assessment:** Nuclei and MN were prepared using the *in vitro* MicroFlow® kit (Litron Laboratories, USA) and analyzed with a FACSCanto II flow cytometer with an integrated high-throughput sampler (Becton Dickinson, USA). Concomitant absolute nuclei counts were made at the start and end of treatment (24 h) or end of the treatment-recovery period (4 h +S9) and used to calculate the relative population doubling (RPD) cytotoxicity parameter. At least 2000 nuclei were evaluated for each concentration assessed.

- **Genotoxicity evaluation:** A response was defined as genotoxic if there was a concentration-related increase in %MN and at least one concentration induced %MN outside the laboratory's historical solvent control range at RPD ≥40%.

- **Phosphorylated histone analysis:** Following treatment, cells were fixed with Cytofix/Cytoperm™ solution (BD Bioscience, USA), and nuclei were stained with 7-AAD and either the mouse anti-H2A.X Phospho (Ser139) FITC-conjugated antibody (Biolegend, USA) or the rabbit anti-phospho-histone H3 (Ser10) Alexa Fluor® 488-conjugated antibody (Cell Signaling Technology, USA) before analysis via flow cytometry.

- **ToxTracker® analysis:** The six green fluorescent protein (GFP)-based mouse embryonic stem reporter cell lines were exposed to the flavour compounds using 3 h +S9 and 24 h treatment conditions (Hendriks et al., 2012). Following treatment/recovery periods, intracellular GFP-associated fluorescence was quantified via flow cytometry. Greater than two-fold induction in GFP fluorescence relative to solvent-treated controls was considered as a positive signal in the assay.

Results (Part 1)

Figure 1. In general, all six NLs increased the pH of the cell culture medium by ~1 unit.

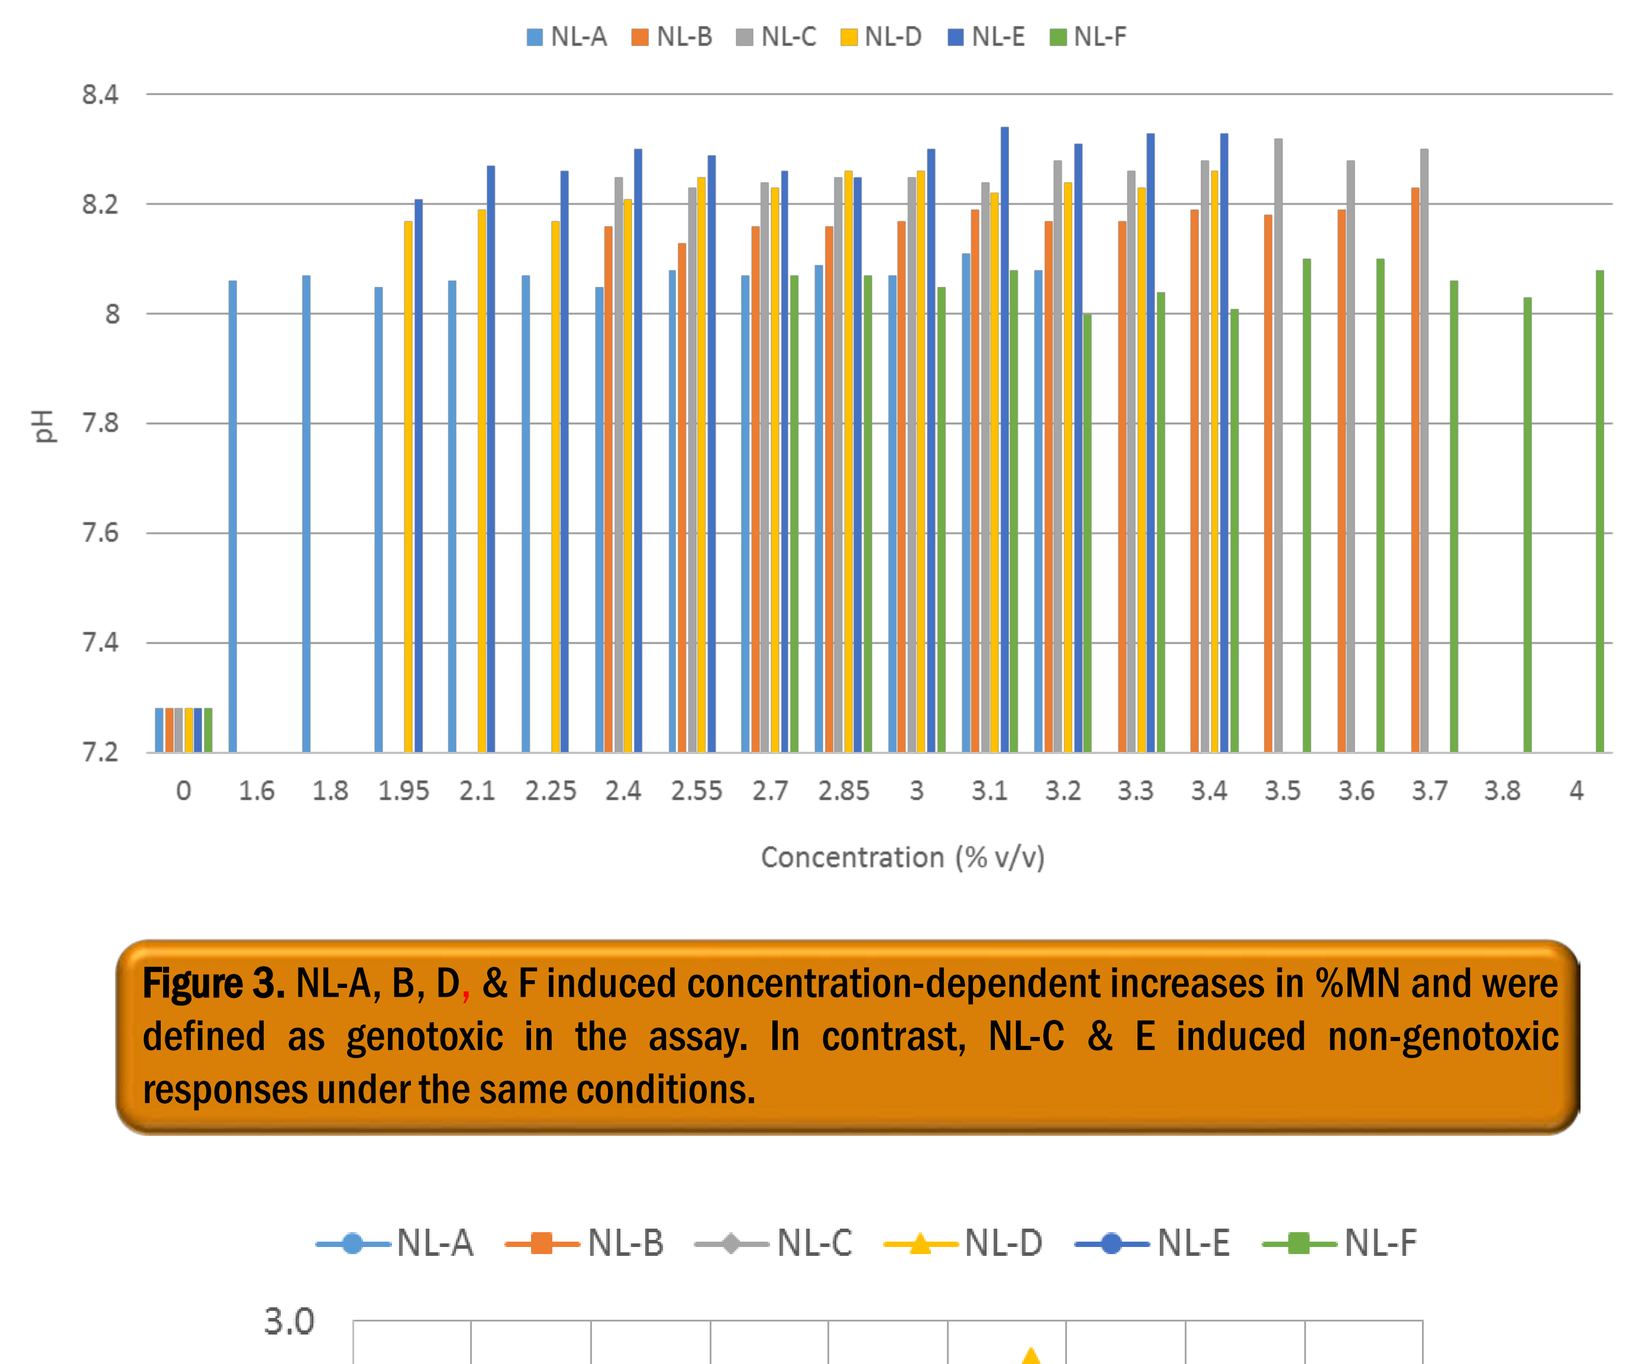
NL-A NL-B NL-C NL-D NL-E NL-F
pH
Concentration (% v/v)

Figure 2. All six NLs increased the osmolality of the cell culture medium in a concentration-dependent manner.

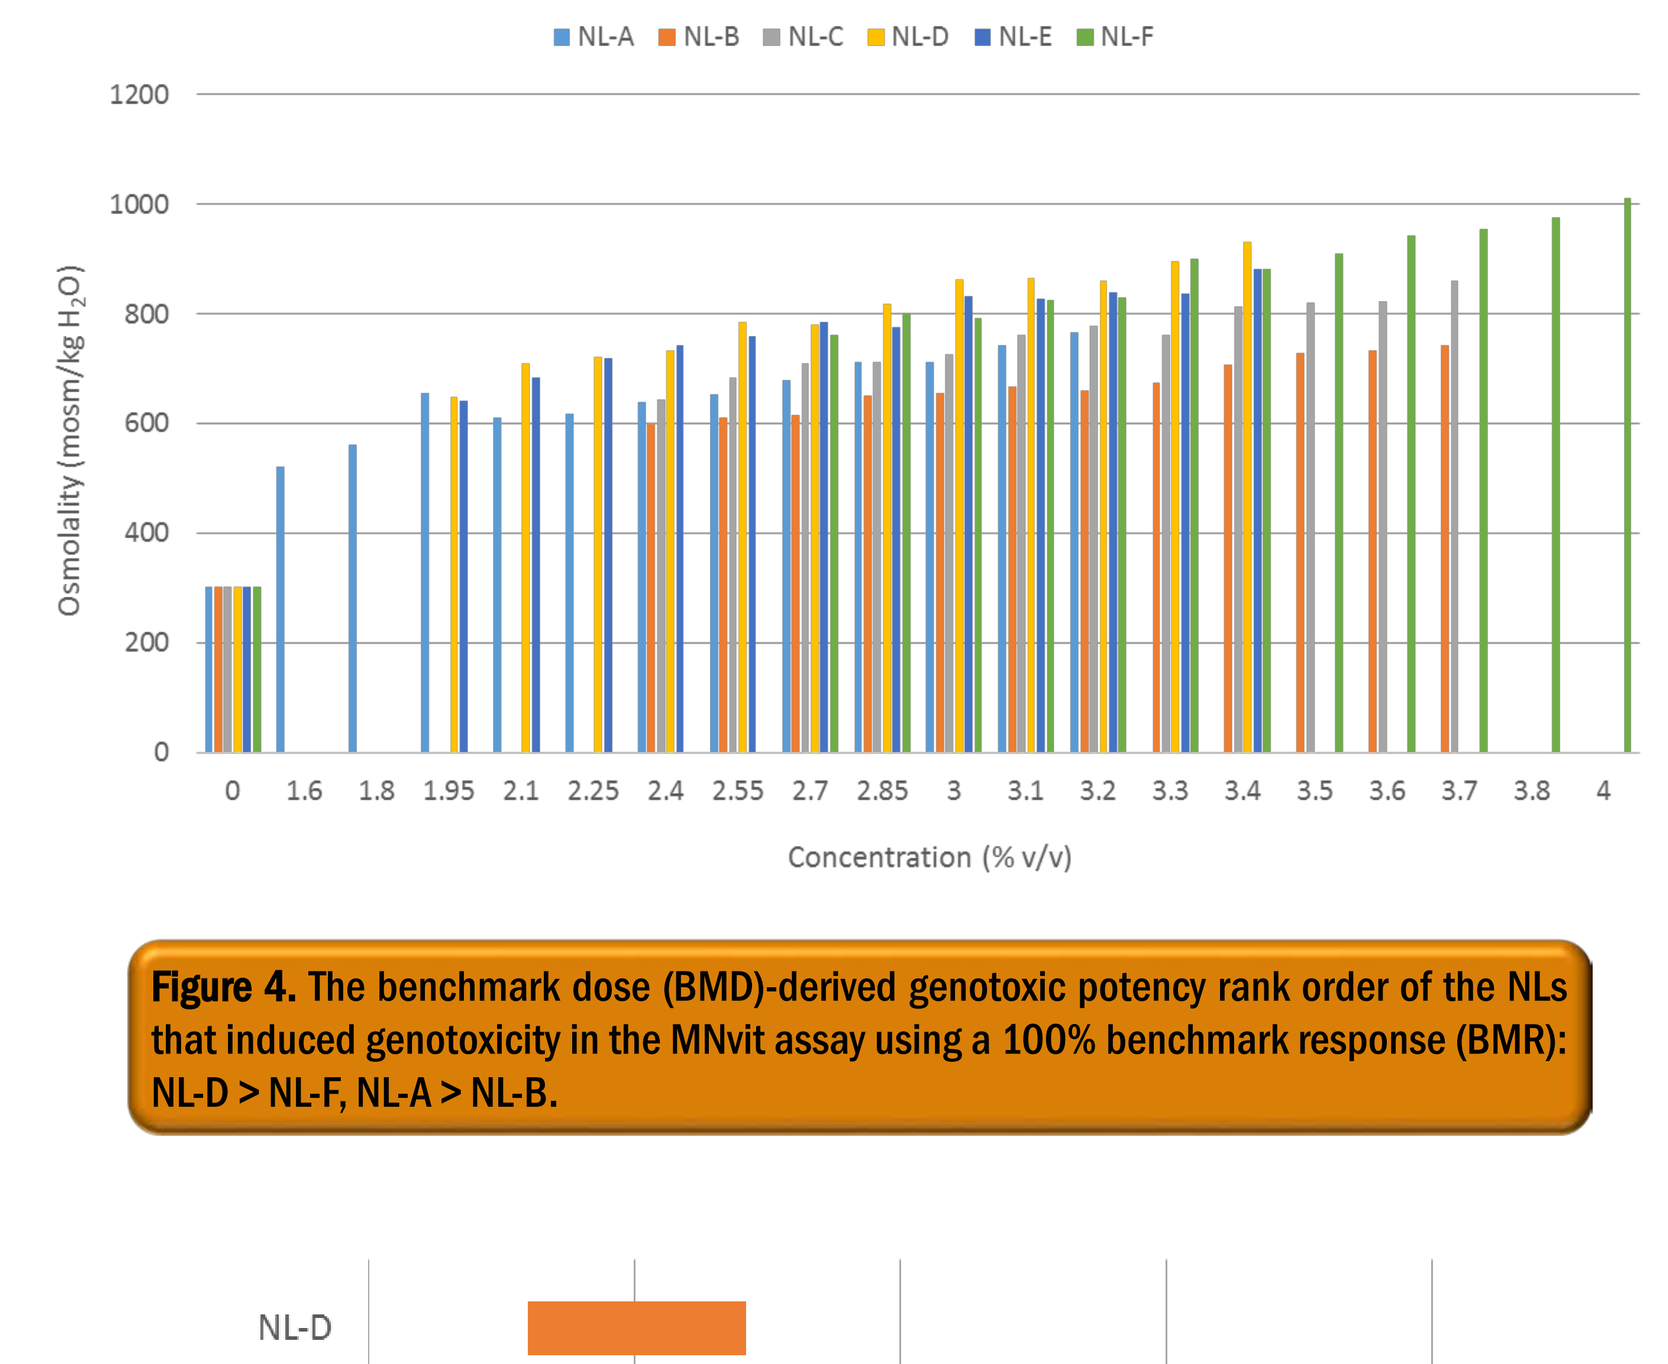
NL-A NL-B NL-C NL-D NL-E NL-F
Osmolality (mosm/kg H2O)
Concentration (% v/v)

Figure 3. NL-A, B, D, & F induced concentration-dependent increases in %MN and were defined as genotoxic in the assay. In contrast, NL-C & E induced non-genotoxic responses under the same conditions.

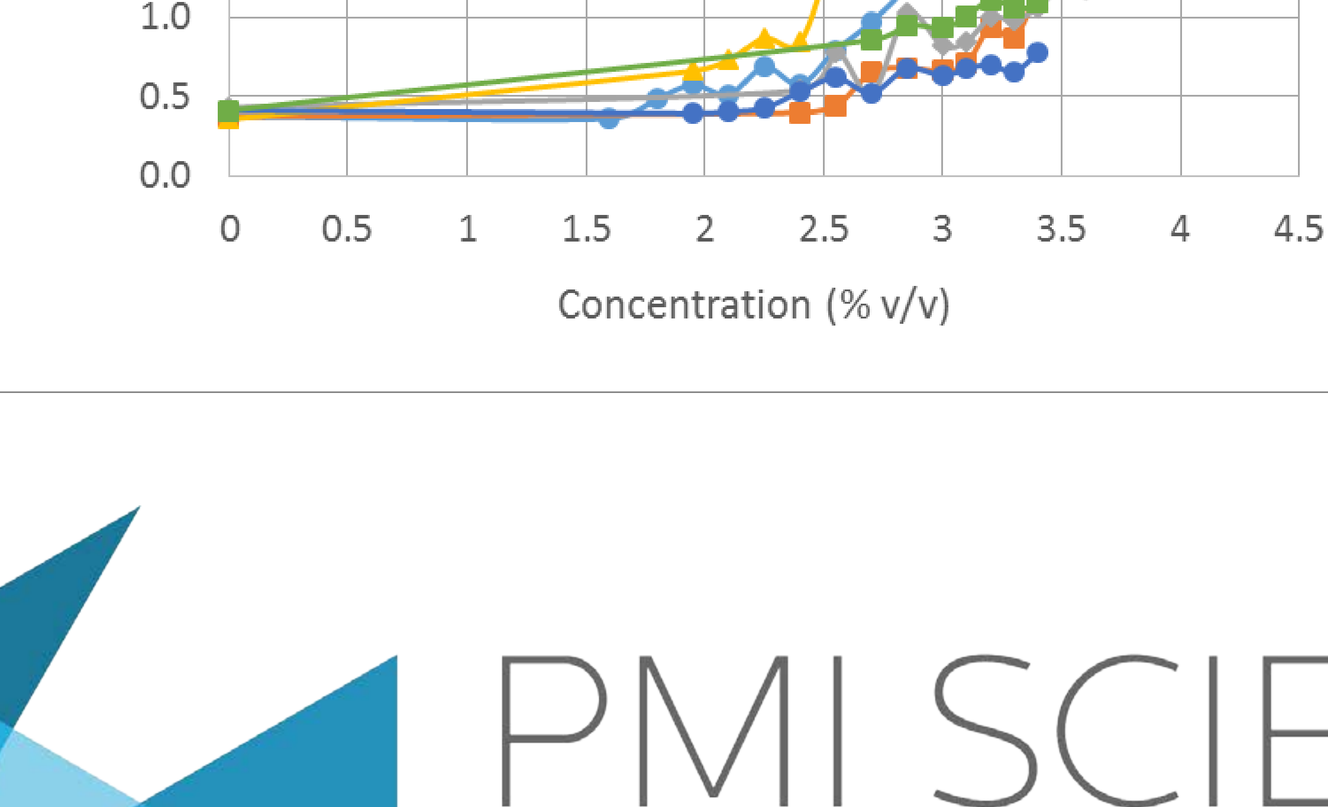
NL-A NL-B NL-C NL-D NL-E NL-F
%MN
Concentration (% v/v)

Figure 4. The benchmark dose (BMD)-derived genotoxic potency rank order of the NLs that induced genotoxicity in the MNvit assay using a 100% benchmark response (BMR): NL-D > NL-F, NL-A > NL-B.

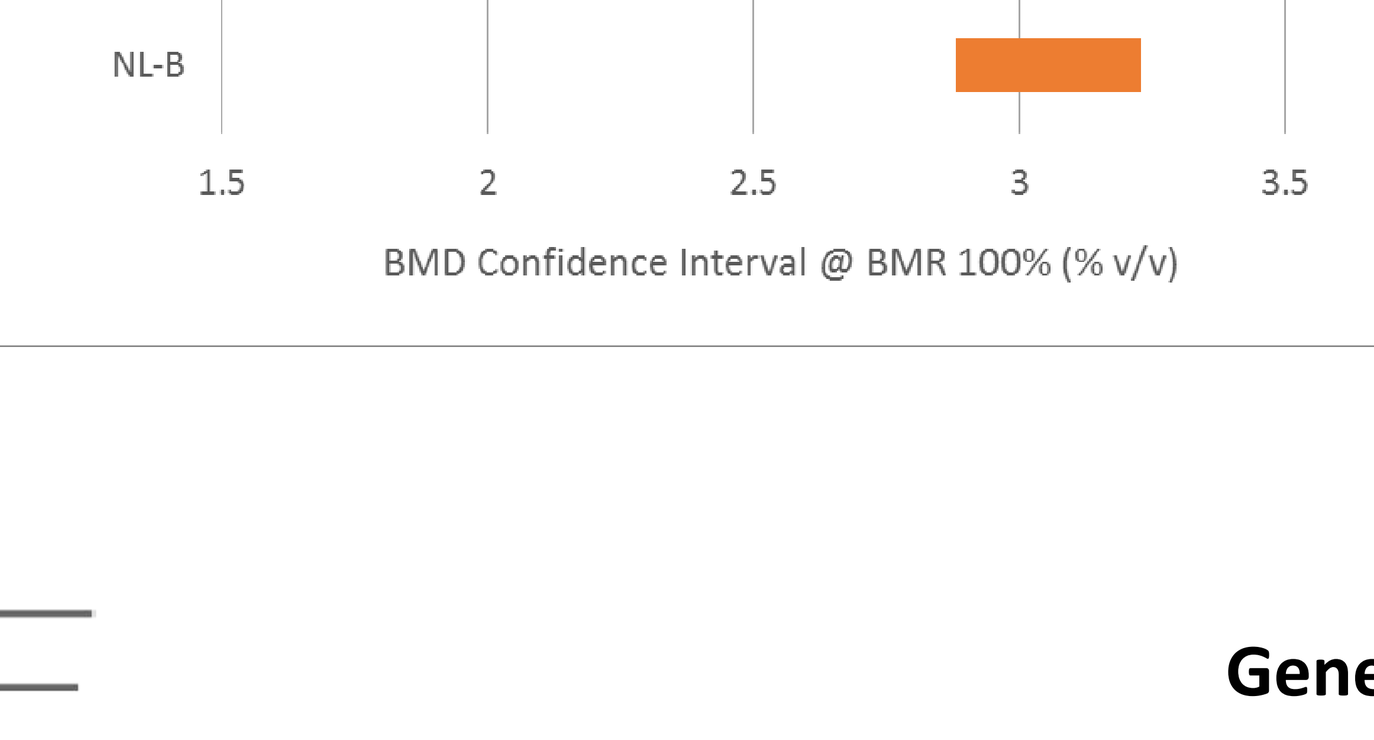
NL-D
NL-F
NL-A
NL-B
BMD Confidence Interval @ BMR 100% (% v/v)

Results (Part 1 cont. and Part 2)

Figure 5. Representative cell cycle histograms of cells exposed to NL-A, NL-D and NL-E for 24 h; negligible evidence of cell cycle arrest even at and beyond the assay's limit of cytotoxicity (RPD 40%).

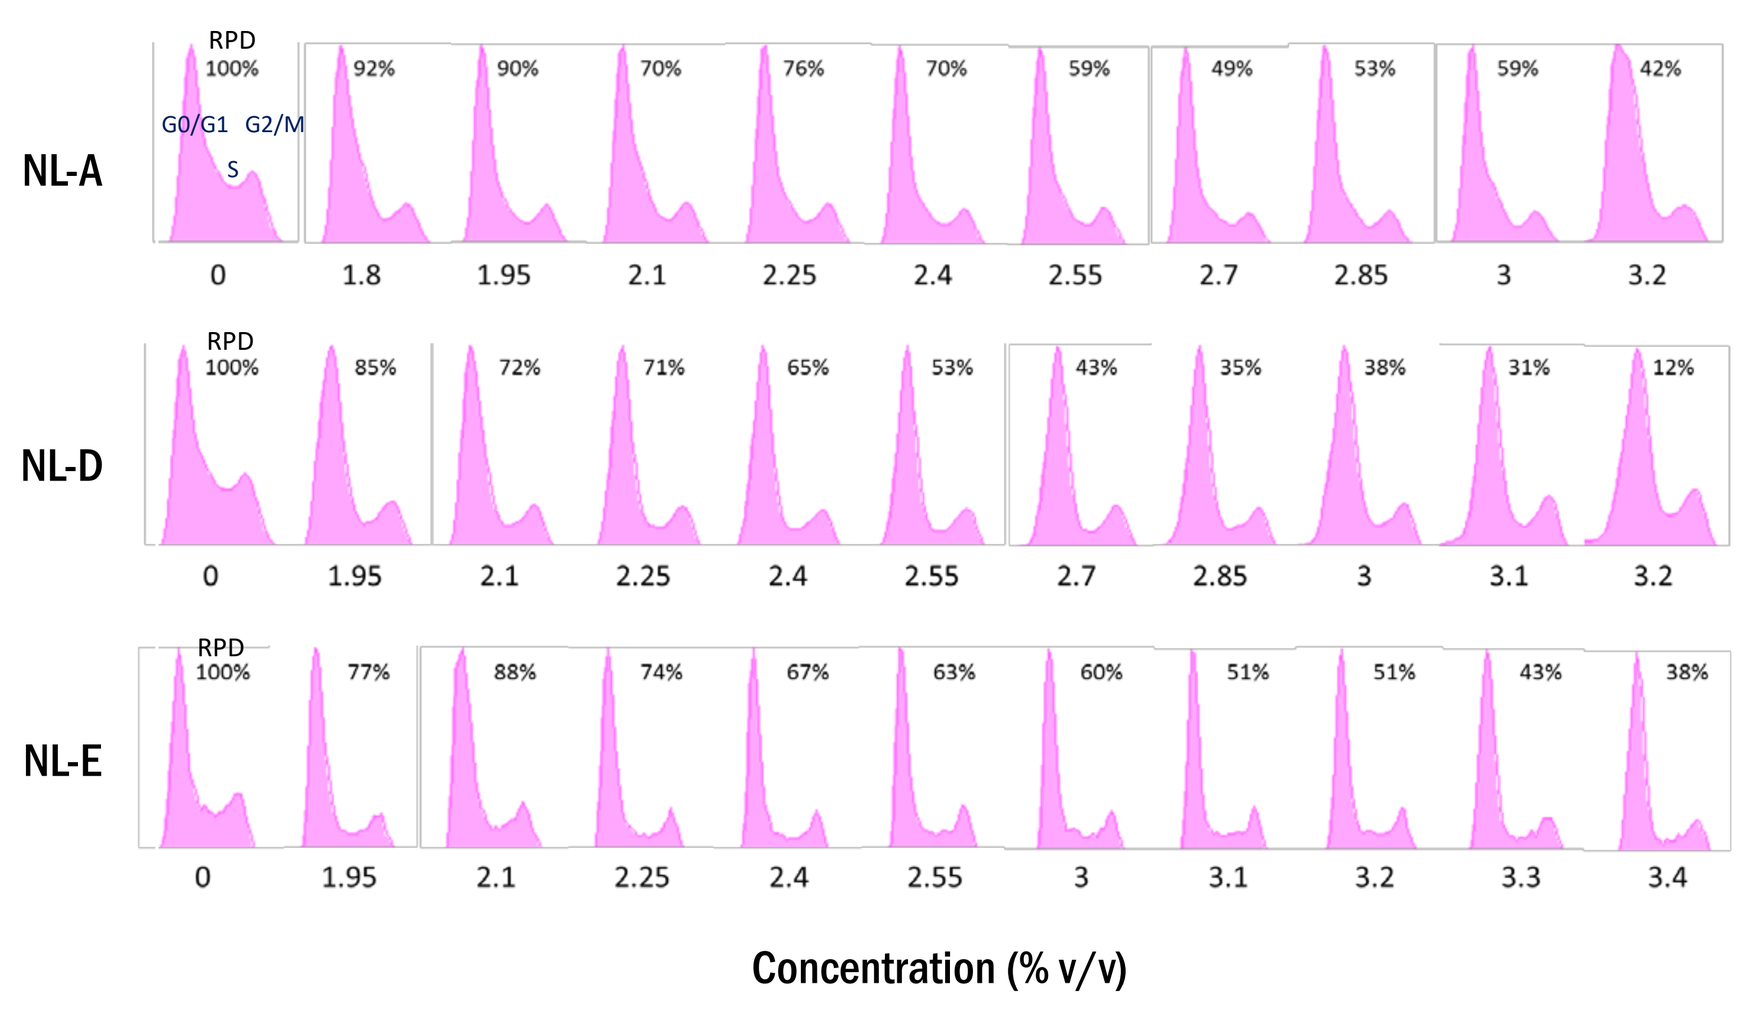
NL-A
NL-D
NL-E
RPD 100%
G0/G1 G2/M
S
Concentration (% v/v)

Figure 6. Negligible changes in the levels of γH2AX and phospho-serine[10]-H3 in cells exposed to NLs for 24 h.

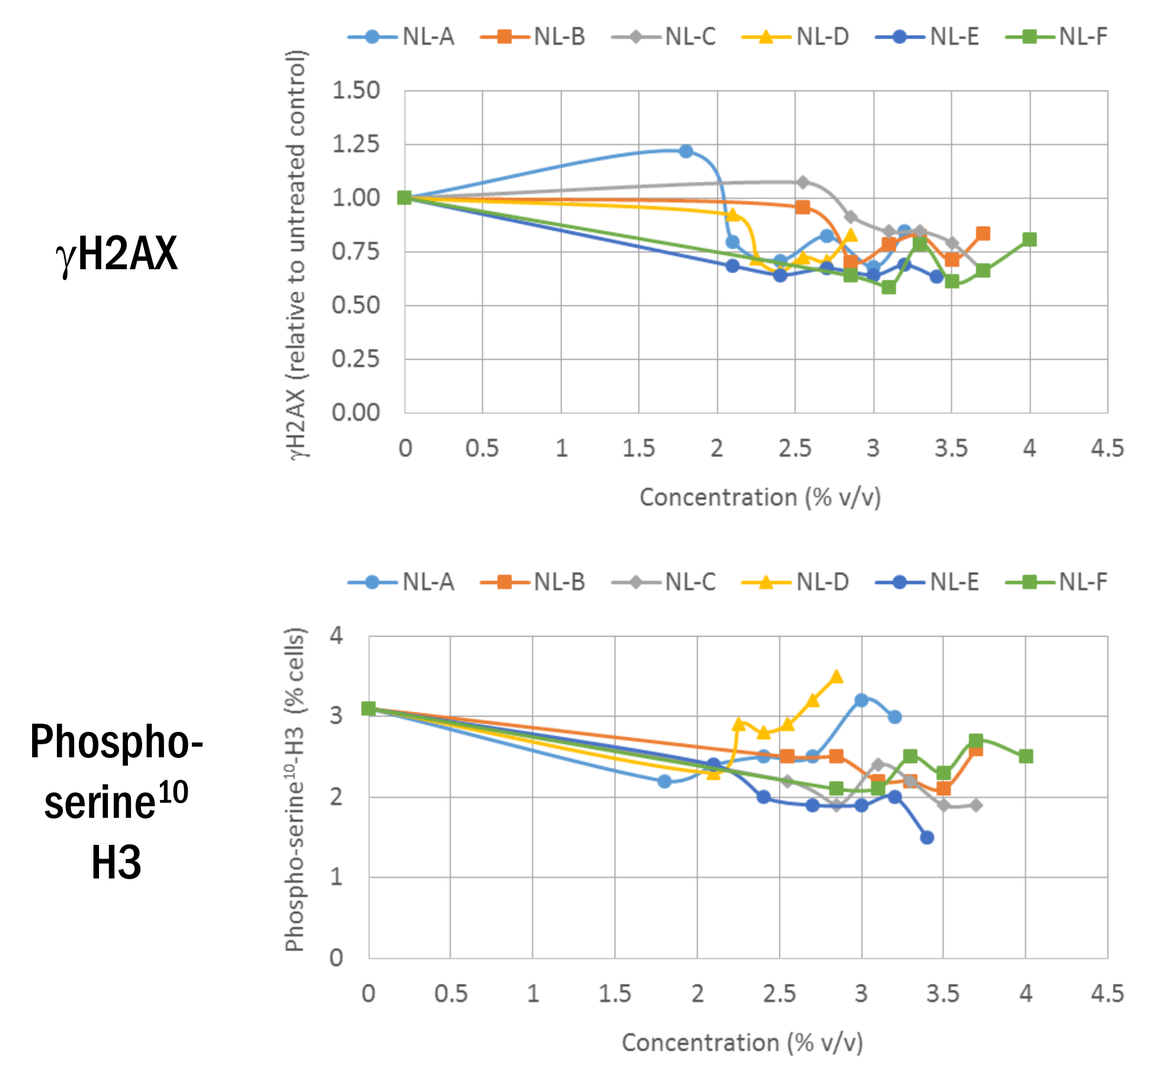
γH2AX
Phospho-serine10 H3
NL-A NL-B NL-C NL-D NL-E NL-F
Concentration (% v/v)

Figure 7. A heat map illustrating the genotoxic potential of the flavour compounds in the two treatment conditions conducted in the MNvit assay: orange (genotoxic) and light yellow (non-genotoxic).

	4 h +S9	24 h
Furaneol		A
Trans-2-hexenal		
Cinnamaldehyde		
Maltol		
Eugenol		
Methyl eugenol		
Cyclohexanone		B
Vanillin		
4-(4-Hydroxyphenyl)-2-butanone	C	
Ethyl n-butyrate		
Ethyl 2-methylbutyrate		

A: Only genotoxic at concentrations 1-10 mM.
B: Potential misleading genotoxic result (see Figure 8)
C: Only genotoxic at concentrations 1-2 mM.

Figure 8. Evidence of G_2M arrest & endoreduplication in cells that exhibited genotoxicity, e.g. in response to trans-2-hexenal and cinnamaldehyde, but not in response to cyclohexanone.

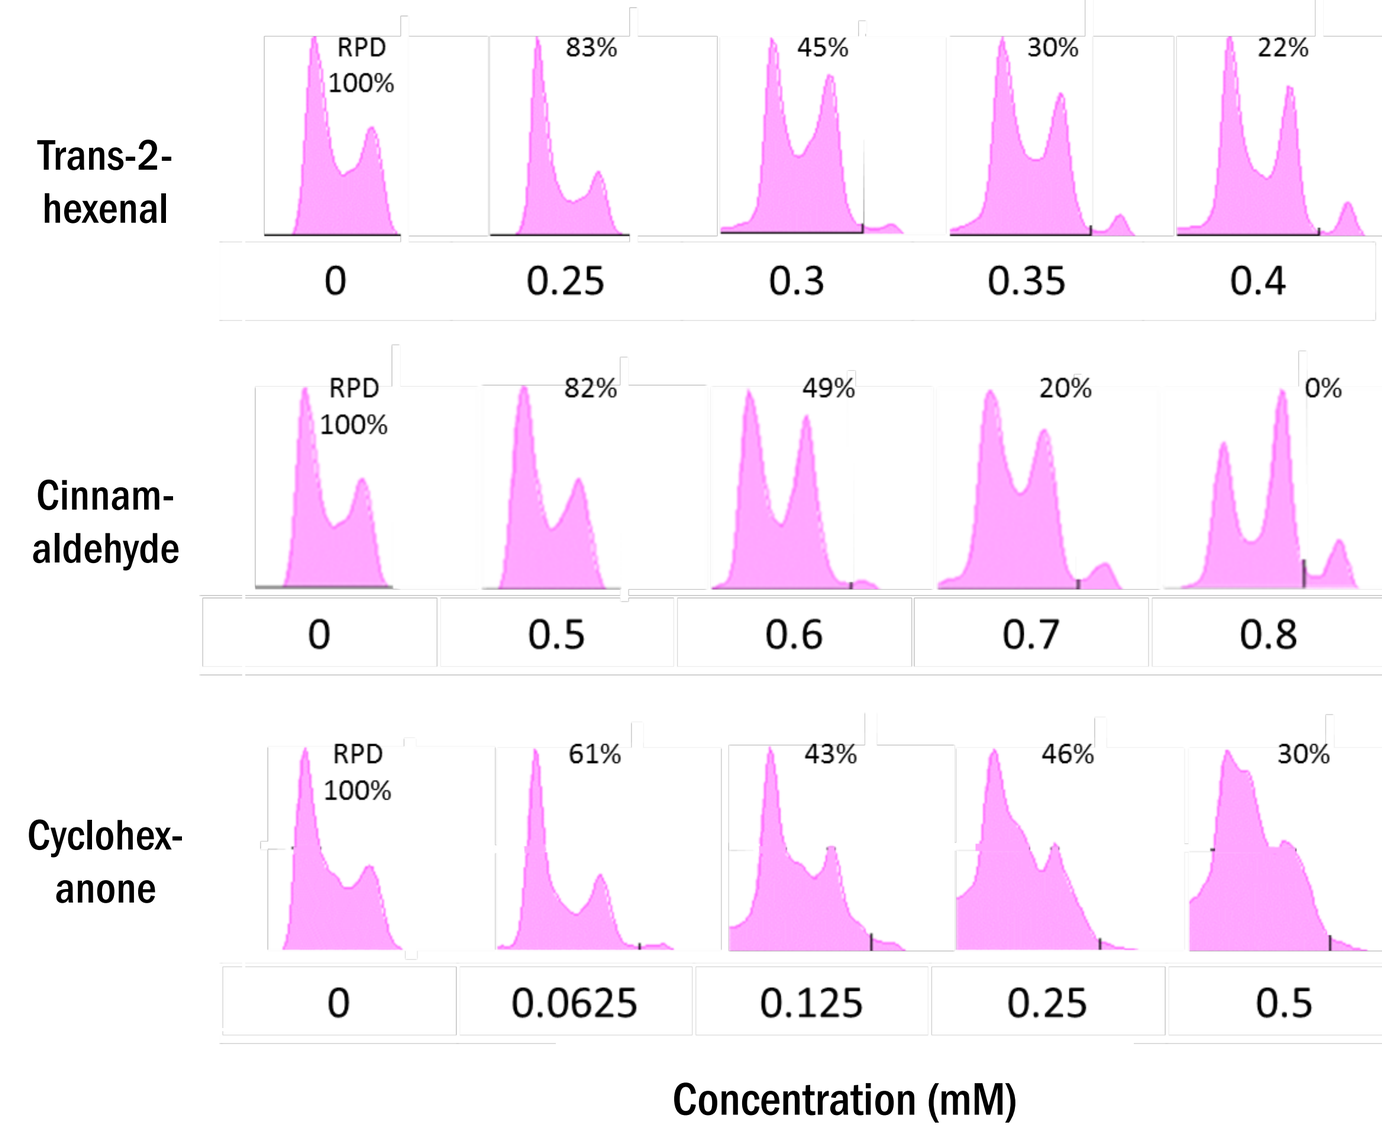
Trans-2-hexenal
Cinnam-aldehyde
Cyclohex-anone
RPD 100%
Concentration (mM)

Figure 9. ToxTracker® showed good overall concordance with the findings from the MNvit assay as well as data from the literature.

Flavour	MNvit	Toxtracker (DNA damage)	Toxtracker (Other)	Literature
Ethyl n-butyrate	–	–	–	Unknown
Ethyl 2-methylbutyrate	–	–	–	Unknown
Vanillin	–	–	+	–
Methyl eugenol	–	–	+	+ DNA adducts
Maltol	–	+ Rtkn	+	+ Ames test
Eugenol	–	+ Rtkn	+	+ Mouse lymphoma assay
Furaneol	+	+	+	+
Trans-2-hexenal	+	+	+	+
Cinnamaldehyde	+	+	+	+
4-(4-Hydroxyphenyl)-2-butanone	+	+	+	Unknown

+ : Positive signal; + Rtkn : Positive signal in Rhotekin DNA damage endpoint; – : Negative signal.

Figure 10. The oxidative stress endpoints (Srnx1 & Blvrb) were prominently activated in response to flavours thought to exert their genotoxicity via an oxidative stress mode-of-action.

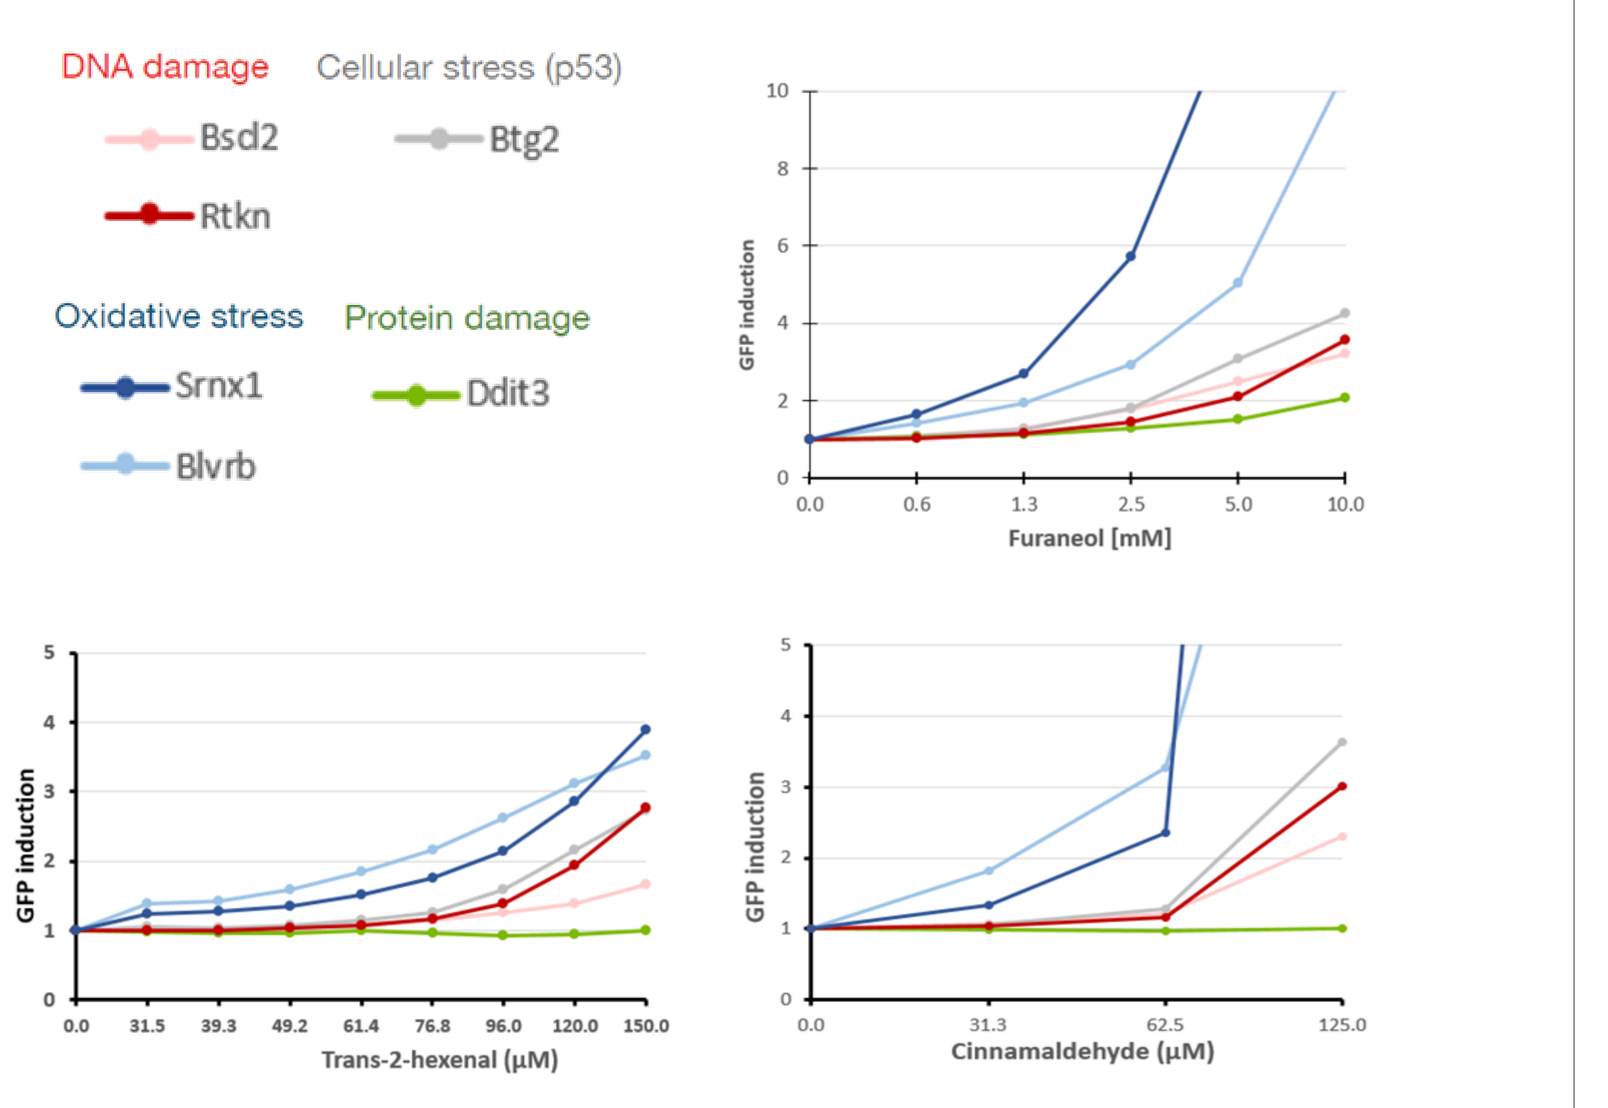
DNA damage
Bsd2
Rtkn
Cellular stress (p53)
Btg2
Oxidative stress
Srnx1
Blvrb
Protein damage
Ddit3
GFP induction
Furaneol [mM]
Trans-2-hexenal (µM)
Cinnamaldehyde (µM)

Discussion and Conclusions

- The four NLs that induced genotoxicity in the MNvit assay all contained PG levels at 40% or more. Furthermore, genotoxic potency was correlated with the level of PG contained within the NL: 100PG > 70PG:20VG > 40PG:40VG. NLs mainly composed of VG, i.e. 20PG:73VG and 100VG, were non-genotoxic in the assay. Taken together, these data corroborate previous reports of PG-induced genotoxicity *in vitro* (Scott *et al.,* 1991).

- Extreme cell culture conditions, i.e. changes in pH and osmolality, *per se* were unlikely to be responsible for the genotoxicity observed as all six NLs produced similar effects on these two parameters.

- Although NL-A, B, D and F induced the formation of MN in the assay, no detectable changes in γH2AX and phospho-serine[10]-H3 biomarkers were observed, and similarly, there was no evidence of cell cycle arrest. These conclusive findings on the advanced genotoxicity endpoints may cast doubt on the relevance of the related MN data.

- Flavours with known *in vitro* genotoxic potential, namely furaneol, trans-2-hexenal and cinnamaldehyde, induced genotoxic responses in the MNvit assay together with G_2M cell cycle phase arrest (and endoreduplication); this signature appears to be indicative of genuine genotoxicity. Based on this assessment, 4-(4-hydroxyphenyl)-2-butanone is likely to be genotoxic *in vitro* upon metabolic activation. In contrast, MN induction in the absence of such cell cycle changes, e.g. as with cyclohexanone, may be a misleading genotoxicity result.

- The other flavours were universally non-genotoxic in the assay. Although maltol (Ames test), eugenol (mouse lymphoma assay) and methyl eugenol (DNA adducts) have been shown to possess genotoxic potential *in vitro*, CHO-WBL cells in combination with induced rat liver S9 in this assay may not be sufficient to produce and detect some DNA lesions (EFSA 2009, Tennant *et al.,* 1987, Myhr & Caspary, 1991, Stening *et al.,* 1997, Randerath *et al.,* 1984 and Phillips *et al.,* 1984).

- Overall, ToxTracker® reliably predicted the genotoxic potential of this subset of flavour compounds relative to MNvit assay and literature findings. The oxidative stress-related endpoints in the assay may be particularly useful to help characterise the genotoxic mode-of-action of flavours *in vitro*.

References

- *EFSA Journal* 2009 ON-879: 1-27; Hendriks G *et al.,* 2012, *Toxicol. Sci.* 125, 285-298; Myhr BC and Caspary WJ. 1991, *Environ. Mol. Mutagen.* 18, 51-83; OECD Test Guideline 487, 2016; Phillips DW *et al.,* 1984, *Carcinogenesis* 5, 1623-28; Randerath K *et al.,* 1984, *Carcinogenesis* 5, 1613-22; Scott D *et al.,* 1991, *Mutat. Res.* 257, 147-204; Stening P *et al.,* 1997, *Human Exper. Toxicol.* 16, 62; Tennant RW *et al.,* 1987, *Science* 236, 933-41.

PMI SCIENCE
PHILIP MORRIS INTERNATIONAL

Genetic Toxicology Association

Delaware, USA

3-4 May 2018

Competing Financial Interest

All authors are employees of Philip Morris International. Philip Morris International is the sole source of funding and sponsor of this project.