Exposure to Cigarette Smoke and Aerosol from the Candidate Modified Risk Tobacco Product THS2.2 on A/J Mice in a Life-Time Inhalation Study

E.T. Wong[1], K. Luettich[2], E. Guedj[2], R. Dulize[2], C. Nury[2], B. Titz[2], P. Leroy[2], G. Vuillaume[2], A. Elamin[2], N.V. Ivanov[2], E. Veljkovic[1], P. Vanscheeuwijck[2], M. C. Peitsch[2], J. Hoeng[2]

[1] Philip Morris International Research Laboratories Pte. Ltd. (part of Philip Morris International group of companies), Singapore
[2] Philip Morris Products S.A. (part of Philip Morris International group of companies), Neuchatel, Switzerland

Poster Board P-04-05-08
Abstract ID 390

Introduction

Smoking causes many serious diseases including lung cancer and chronic obstructive pulmonary disease (COPD). Mouse models have been employed to study the carcinogenicity of chemical compounds and infer underlying mechanisms of lung tumor development in humans[1]. Different strains of mice display markedly varied sensitivity to lung tumor development. For example, mice of the C57Bl/6 strain are rather resistant to tumor induction, while Balb/c mice are considered intermediate in susceptibility[2]. In contrast, the A/J mouse is highly susceptible to lung tumor induction and has been widely used as a screening system in carcinogenicity testing. Although this mouse strain is developing lung tumors spontaneously, an 18-month inhalation period was previously found to be sufficient in eliciting a CS concentration-dependent lung tumor response[3]. Moreover, gene and microRNA (miRNA) expression analysis demonstrated that lung tumors developing in MS-exposed A/J mice exhibit marked differences compared to spontaneously arising tumors, and that these differences can be harnessed in the form of a gene signature to potentially predict the extent of exposure.[4]

Here, we report on an 18-month *in vivo* inhalation study, performed on A/J mice, to assess the chronic toxicity of exposure to aerosol from THS2.2, a candidate modified risk tobacco product (cMRTP), and to compare it with the toxicity of smoke from the 3R4F reference cigarette (CS). This mouse model has previously been shown to be susceptible to develop lung tumors and has been used in many carcinogenicity studies.

Study Design and Endpoints

Female A/J mice were whole-body exposed to filtered air (Sham), to three concentrations of THS2.2 aerosol (6.7, 13.4, 26.8 µg/L nicotine) and one concentration of 3R4F CS (13.4 µg/L nicotine) for 6 h/day, 5 d/week, for up to 18 months; male mice were exposed to filtered air (Sham) and the highest concentration of THS2.2 aerosol (Figure 1). Care and use of the mice was in accordance with the National Advisory Committee for Laboratory Animal Research Guideline 2004[5]. All animal experiments were approved by the Institutional Animal Care and Use Committee (IACUC). Animals were observed on a daily basis, body weight progression was monitored once per week, exposure parameters were measured at regular intervals. Toxicological assessment was performed according to published standard methods[6, 7] including histopathological evaluation, which is currently still ongoing. Interim dissections were performed after 1, 5 and 10 months of exposure. At months 1 and 5, animals were also allocated for the following endpoints: bronchoalveolar lavage fluid (BALF) analysis by FACS and multi-analyte (cytokines/chemokines) profiling; lung function tests. At each time point, nasal epithelium, trachea, lungs and blood were collected from a subset of animals for an extensive molecular high-throughput analysis (transcriptomics, proteomics).

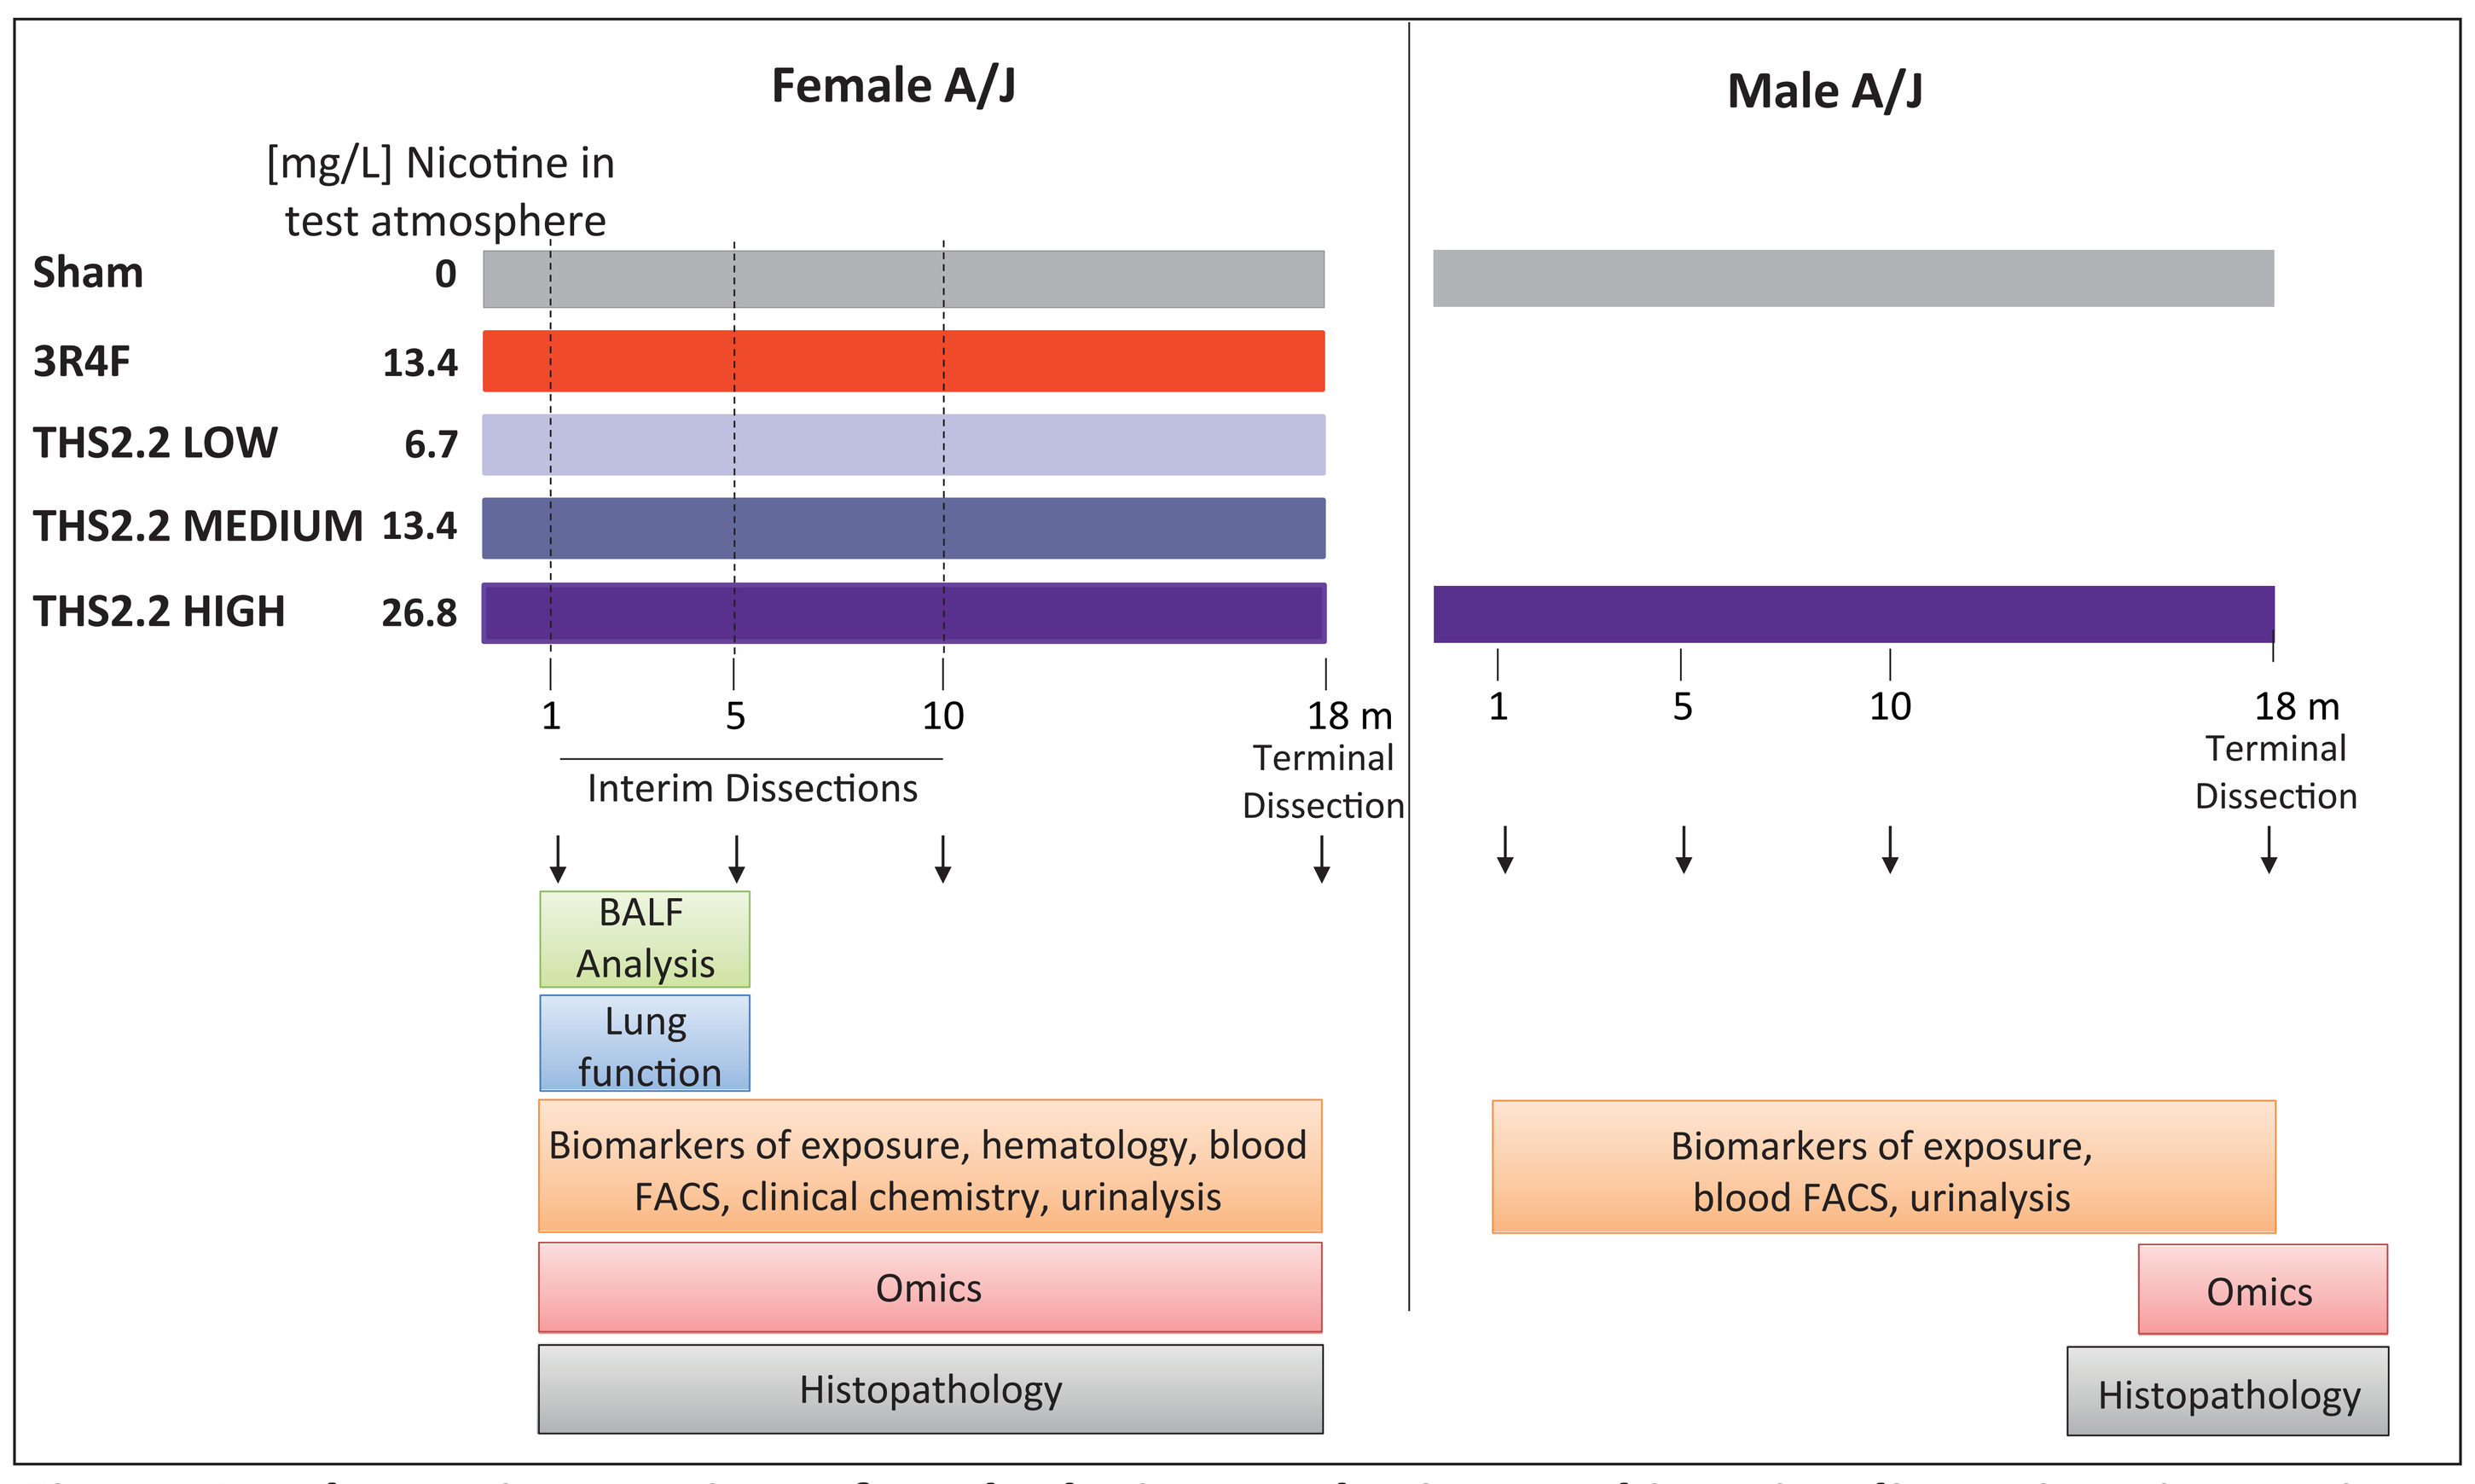
Female A/J
Male A/J
[mg/L] Nicotine in test atmosphere
Sham 0
3R4F 13.4
THS2.2 LOW 6.7
THS2.2 MEDIUM 13.4
THS2.2 HIGH 26.8
1 5 10 18 m
Terminal Dissection
Interim Dissections
BALF Analysis
Lung function
Biomarkers of exposure, hematology, blood FACS, clinical chemistry, urinalysis
Biomarkers of exposure, blood FACS, urinalysis
Omics
Histopathology

Figure 1. Schematic overview of study design, endpoints and interim dissection time points.

Aerosol Exposure and Uptake

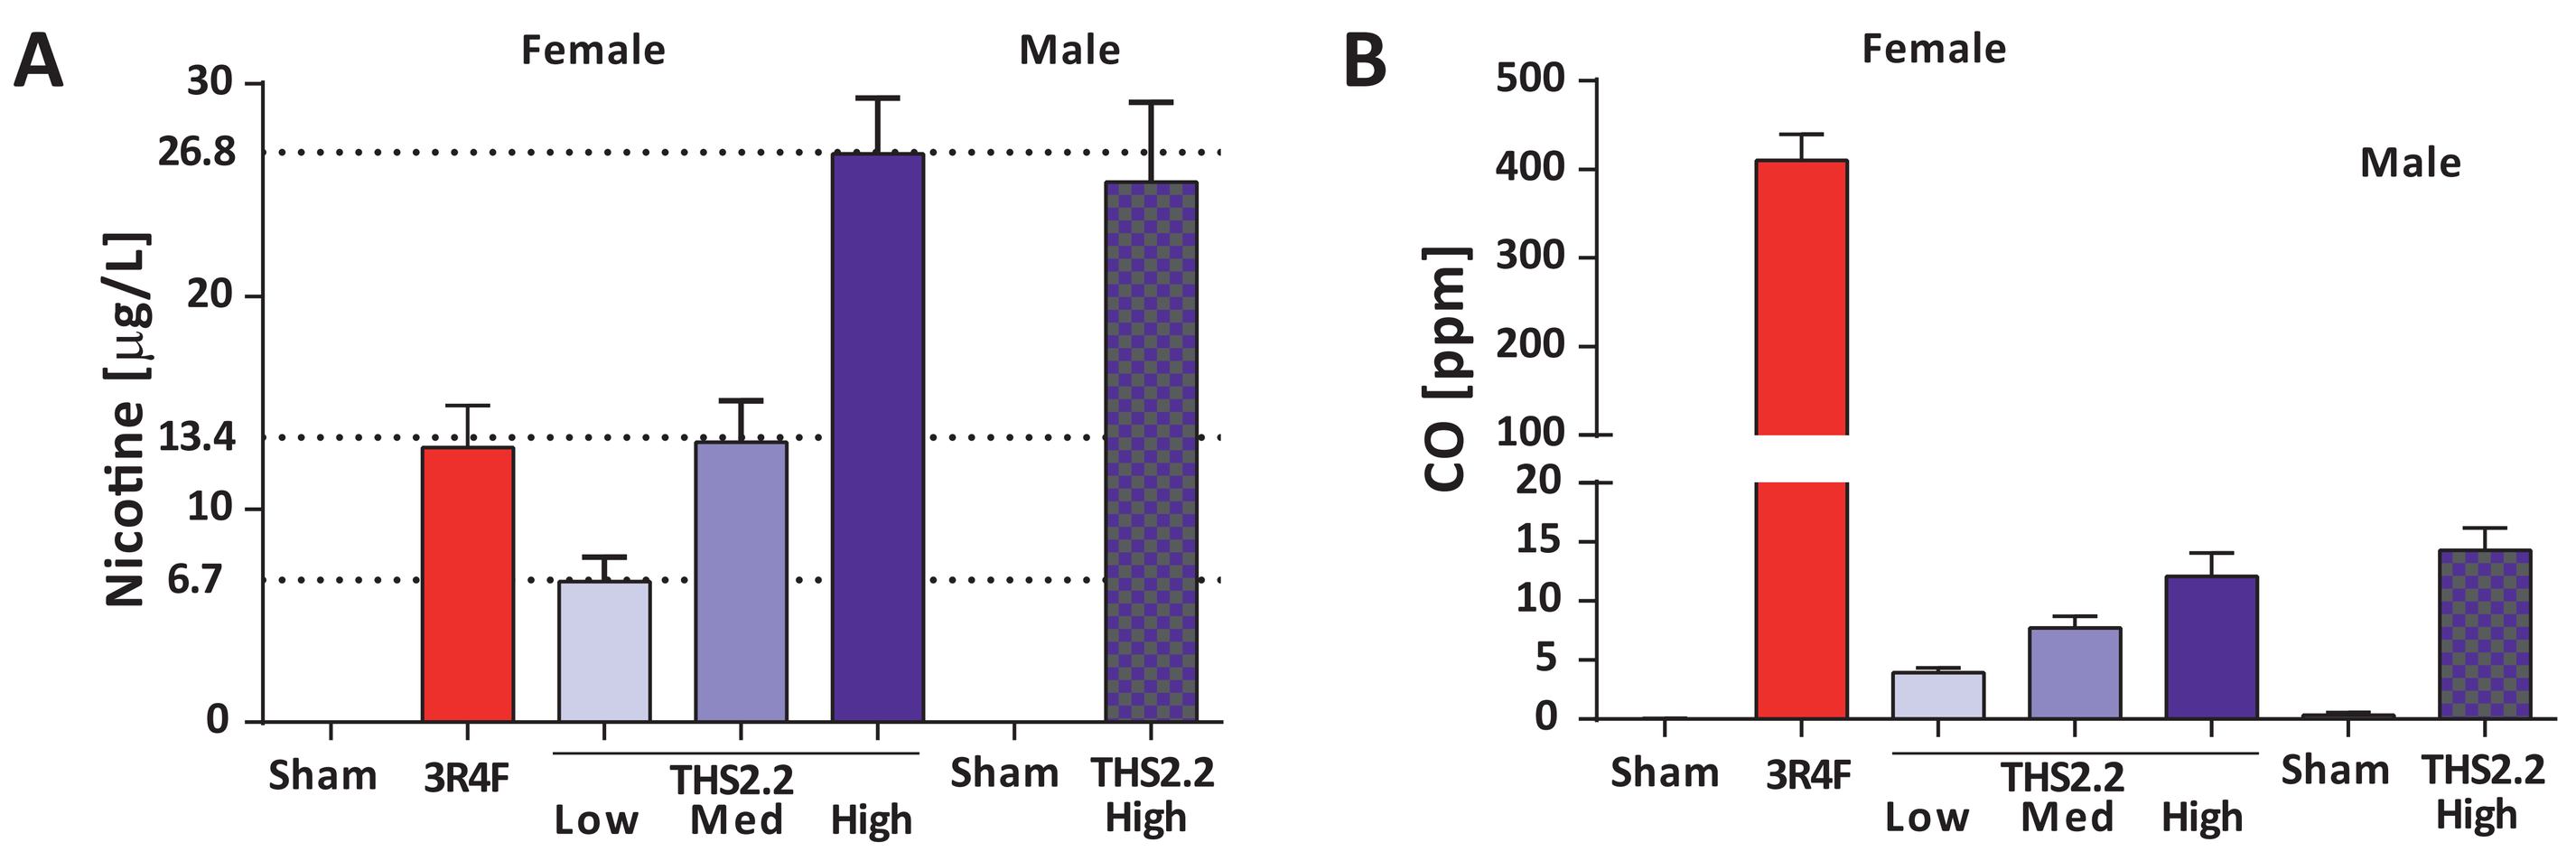
A
Female
Male
Nicotine [µg/L]
Sham 3R4F THS2.2 Low Med High Sham THS2.2 High
B
CO [ppm]

Figure 2. (A) Nicotine and (B) CO concentration in the exposure chambers. *Data are presented as mean ± SD. Dotted lines indicate nicotine target concentrations.*

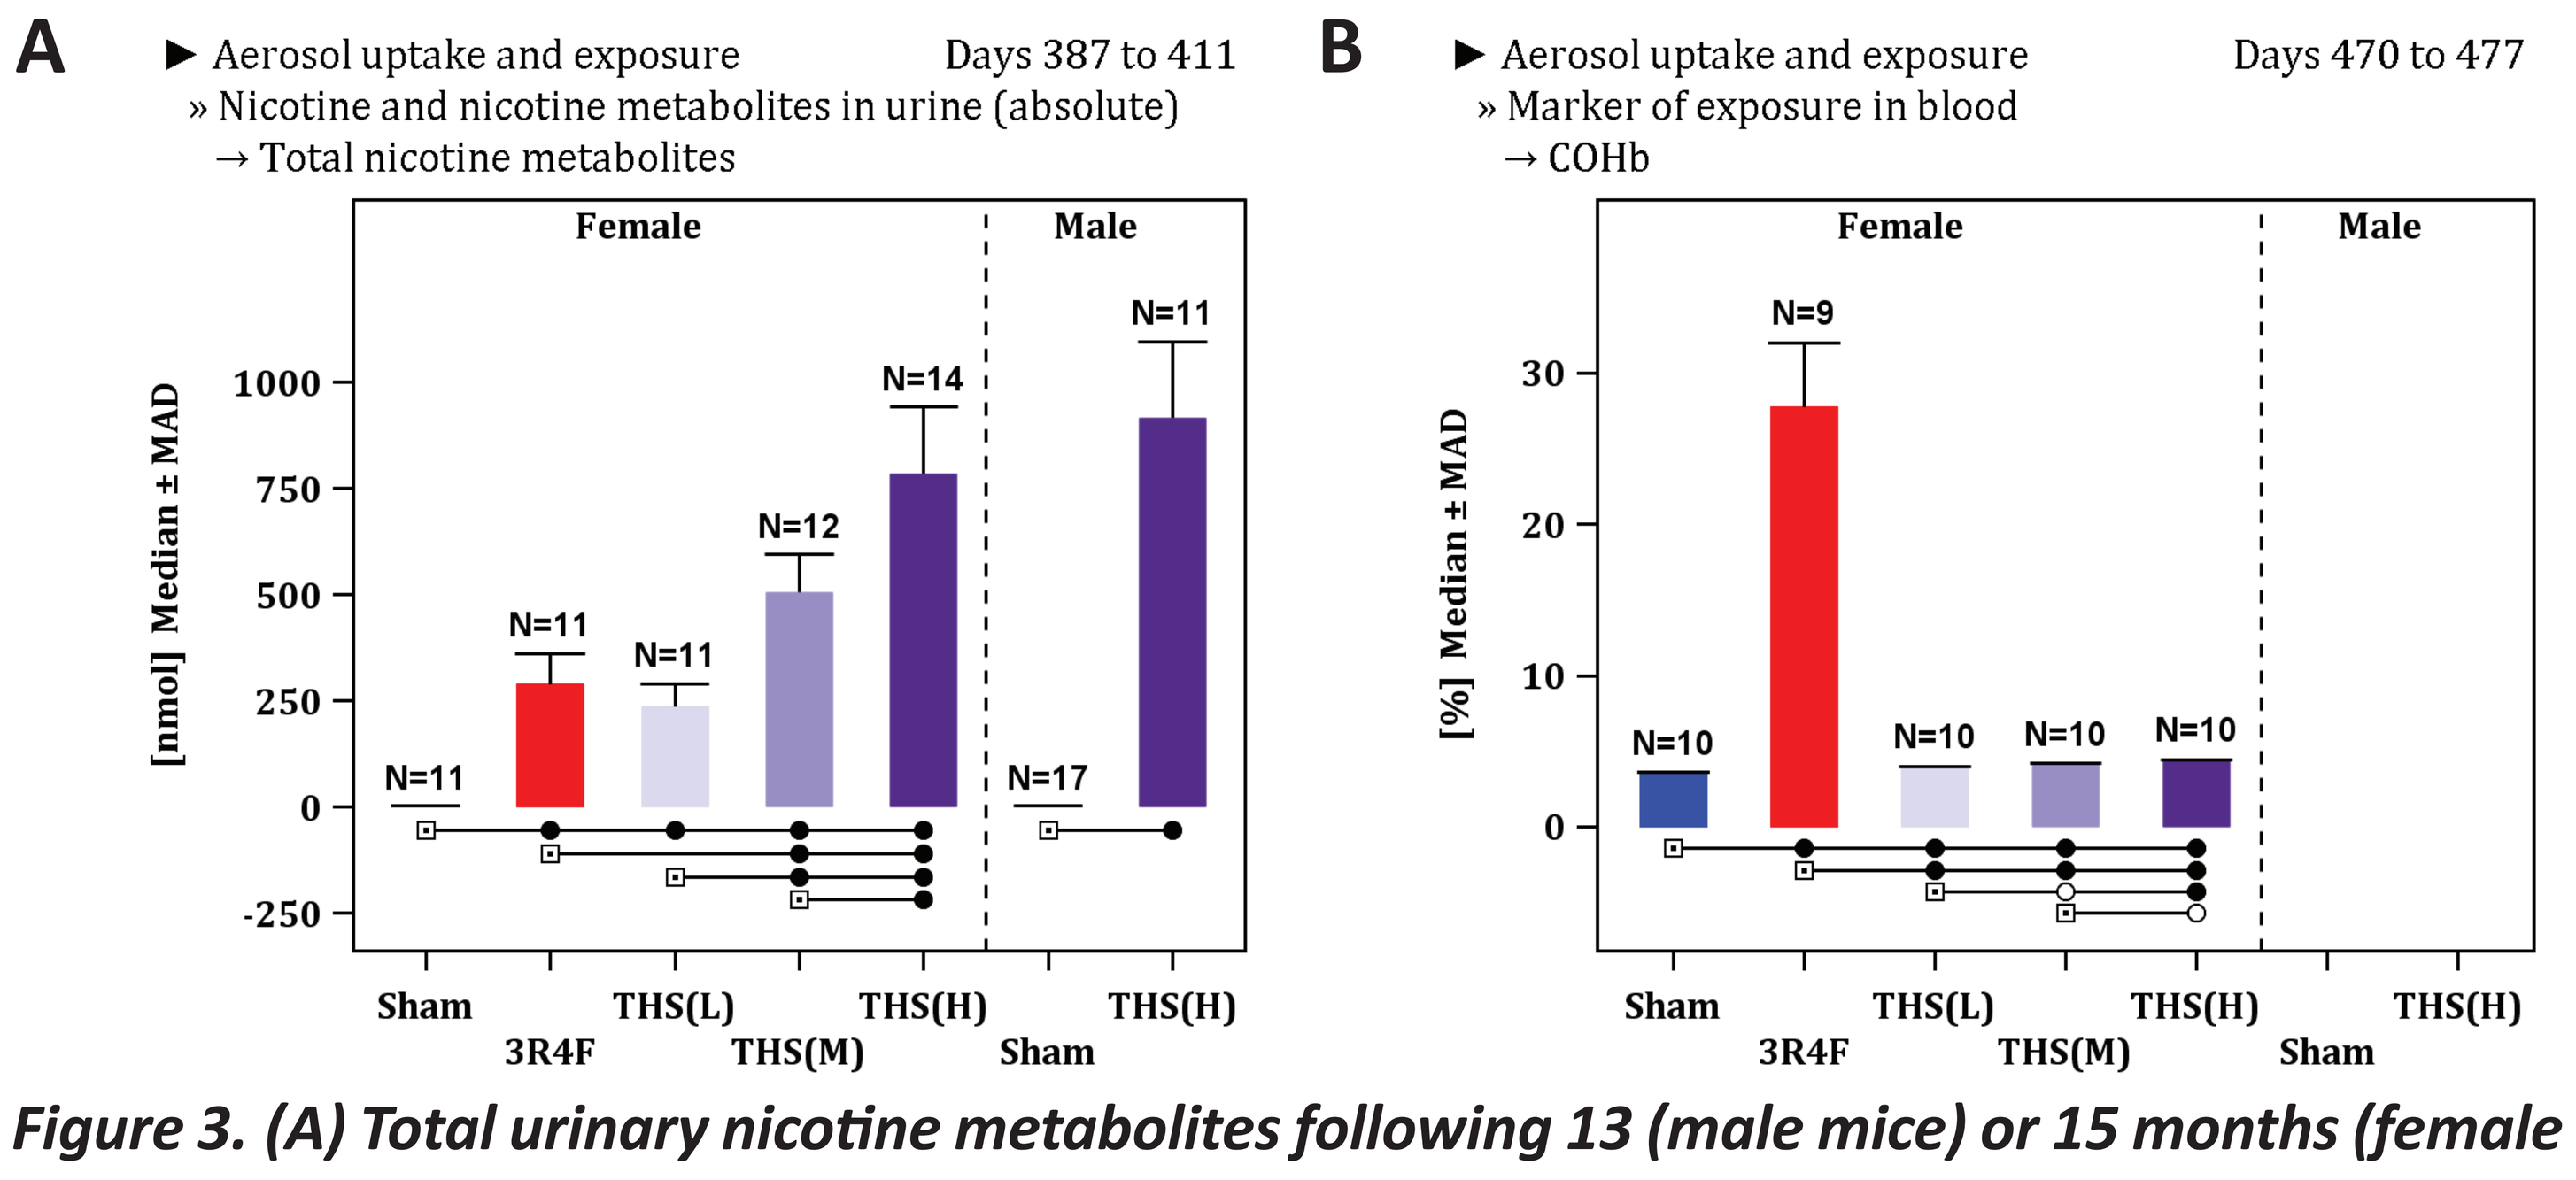
A
► Aerosol uptake and exposure
» Nicotine and nicotine metabolites in urine (absolute)
→ Total nicotine metabolites
Days 387 to 411
[nmol] Median ± MAD
B
► Aerosol uptake and exposure
» Marker of exposure in blood
→ COHb
Days 470 to 477
[%] Median ± MAD

Figure 3. (A) Total urinary nicotine metabolites following 13 (male mice) or 15 months (female mice) of exposure. (B) Blood carboxyhemoglobin levels following 16 months (female mice) of exposure. *Data are presented as mean ± MAD. Lines indicate statistical group comparisons (t-test) where p-values fell below 0.05 (not adjusted).*

References

1) Meuwissen, R & A Berns (2005). Mouse models for human lung cancer. Genes & Dev 19(6):643-664.
2) Gordon, T & M Bosland (2009). Strain-dependent differences in susceptibility to lung cancer in inbred mice exposed to mainstream cigarette smoke. Cancer Letters 275(2):213-220.
3) Stinn, W et al. (2013). Towards the validation of a lung tumorigenesis model with mainstream cigarette smoke inhalation using the A/J mouse. Toxicology 305: 49-64.
4) Luettich, K et al. (2014). Systems toxicology approaches enable mechanistic comparison of spontaneous and cigarette smoke-related lung tumor development in the A/J mouse model. Interdiscip Toxicol 7(2):73-84.
5) National Advisory Committee for Laboratory Animal Research (NACLAR)(2004). Guidelines on the Care and Use of Animals for Scientific Purposes.
6) OECD (2009), Test No. 453: Combined chronic toxicity/ Carcinogenicity Studies.
7) Phillips, B et al. (2016). An 8-month systems toxicology inhalation/ cessation study in Apoe-/- mice to investigate cardiovascular and respiratory exposure effects of a candidate modified risk tobacco product, THS2.2, compared with conventional cigarettes. Toxicol Sci 149(2): 411-432.

General Health, Survival, Hematology and Clinical Chemistry

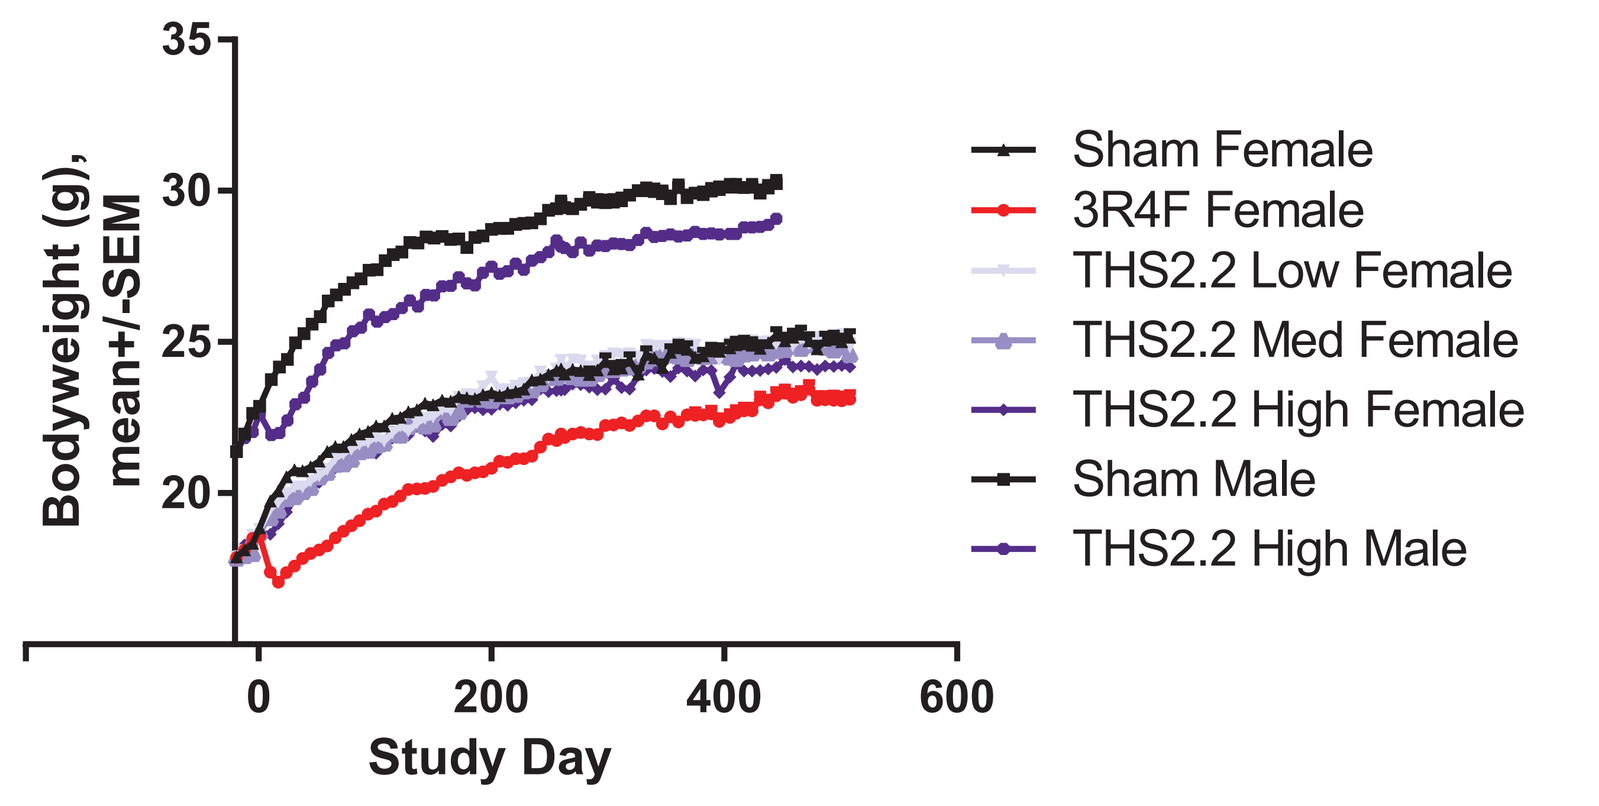
Bodyweight (g), mean+/-SEM
Study Day
Sham Female
3R4F Female
THS2.2 Low Female
THS2.2 Med Female
THS2.2 High Female
Sham Male
THS2.2 High Male

Figure 4. Body weight development throughout the course of the study.

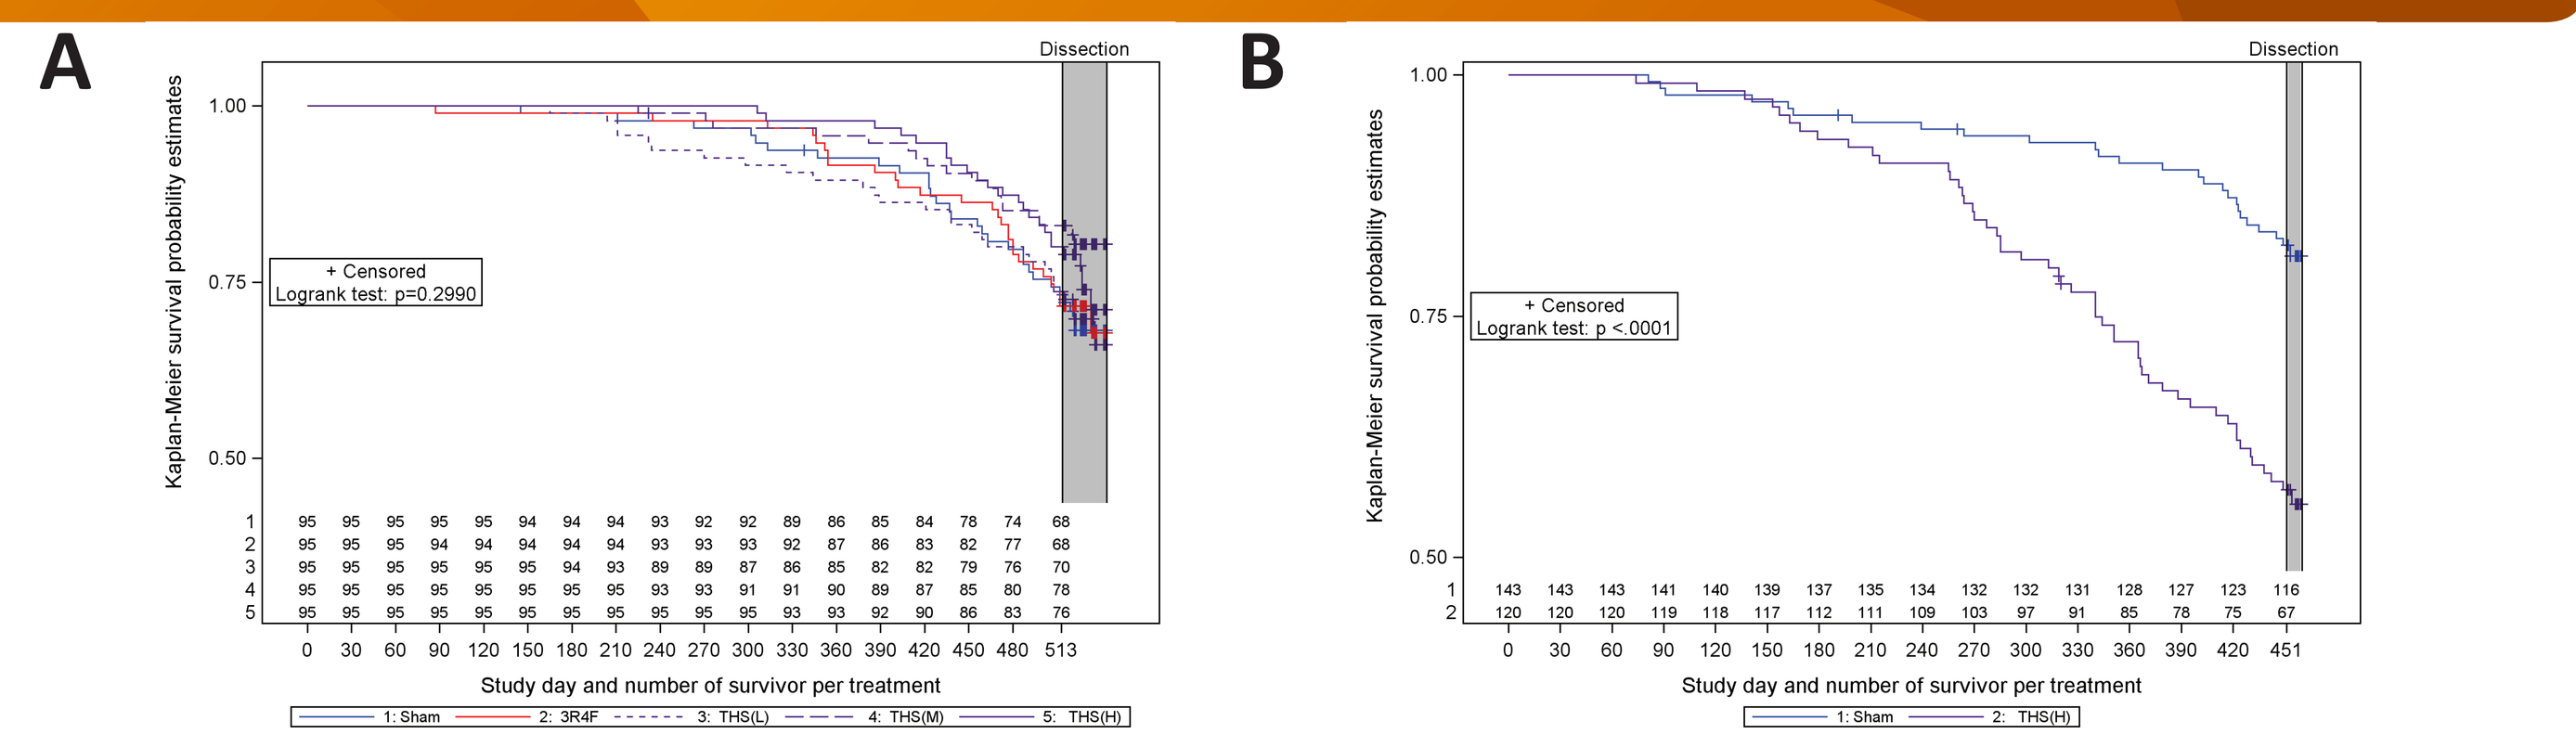
A
B
Dissection
Kaplan-Meier survival probability estimates
+ Censored
Logrank test: p=0.2990
Logrank test: p <.0001
Study day and number of survivor per treatment

Figure 5. Survival statistics for (A) female and (B) male A/J mice. *Data are presented as Kaplan-Meier survival probablility estimates throughout the course of the study. + indicates censored data (technical deaths).*

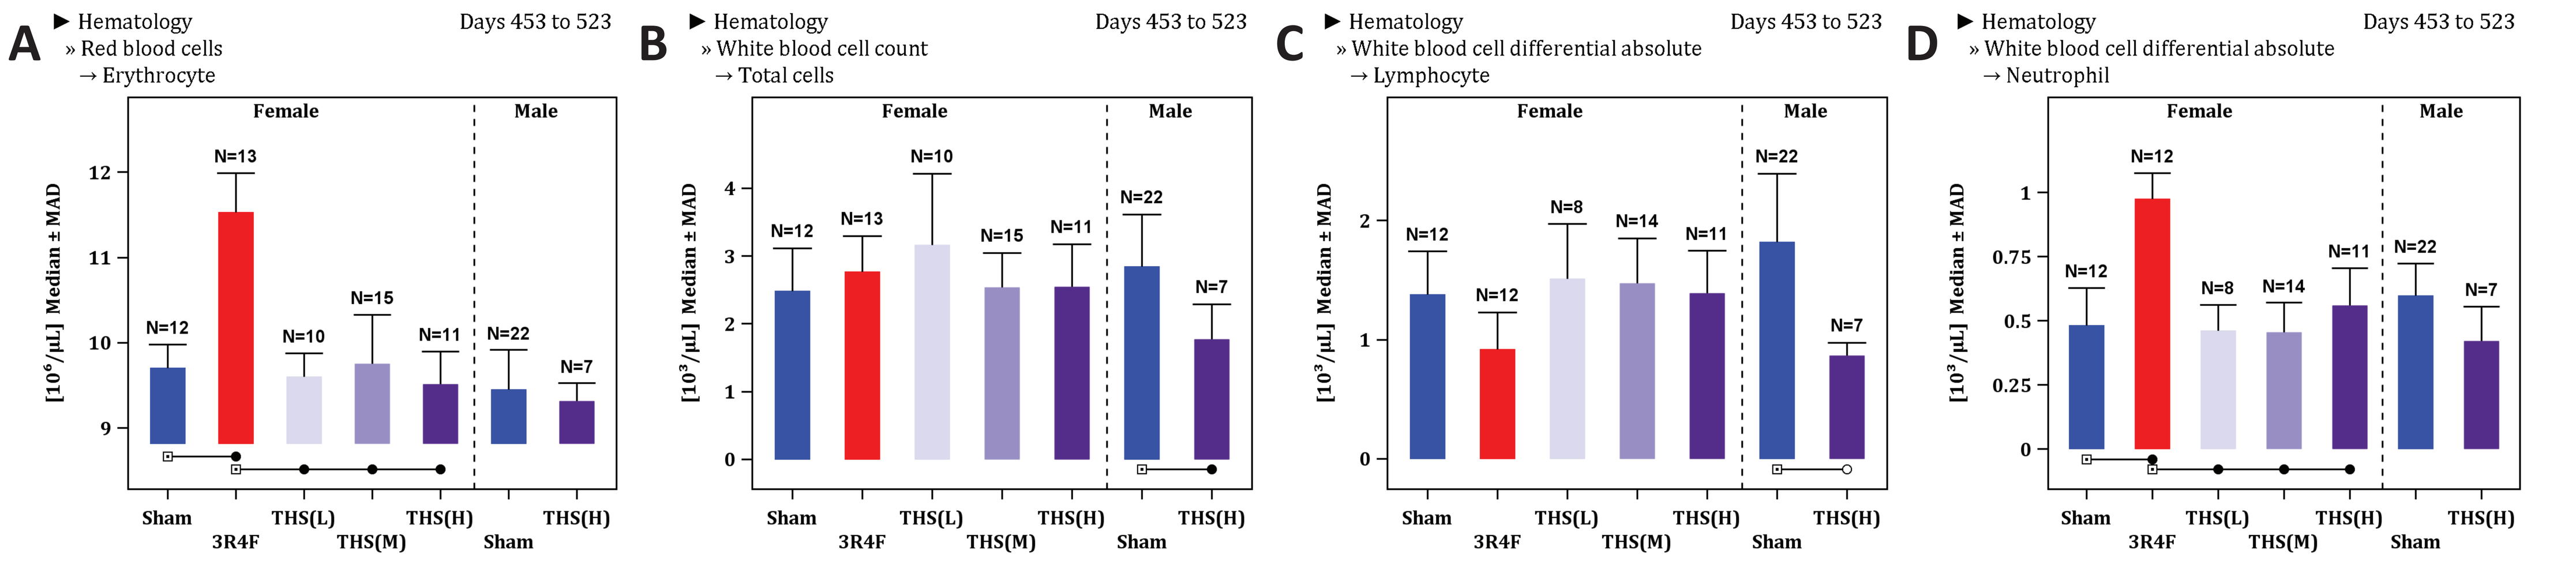
A
► Hematology
» Red blood cells
→ Erythrocyte
Days 453 to 523
B
► Hematology
» White blood cell count
→ Total cells
C
► Hematology
» White blood cell differential absolute
→ Lymphocyte
D
► Hematology
» White blood cell differential absolute
→ Neutrophil
Female
Male
Sham 3R4F THS(L) THS(M) THS(H) Sham THS(H)

Figure 6. (A) Red blood cell (erythrocyte), (B) total white blood cell, (C) absolute lymphocyte and (D) absolute neutrophil counts in blood of in A/J mice following 15 (male mice) or 18 (female mice) months of exposure. *Data are presented as median±mean absolute deviation (MAD). Lines indicate statistical differences between groups (p<0.05, t-test, not adjusted).*

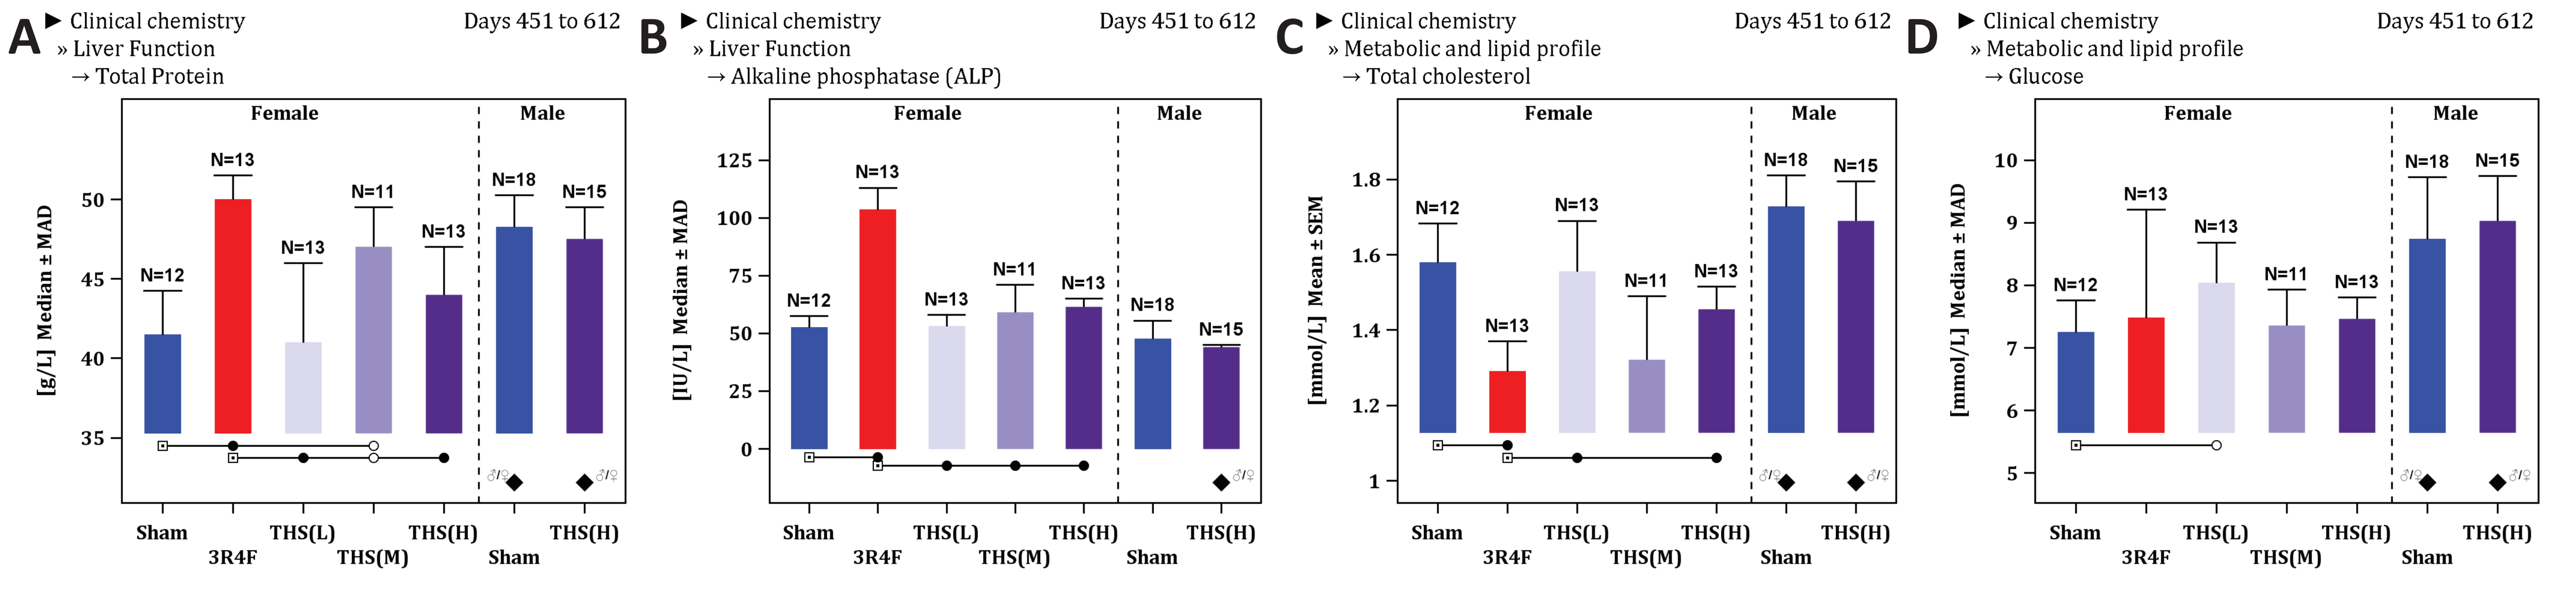
A
► Clinical chemistry
» Liver Function
→ Total Protein
Days 451 to 612
B
► Clinical chemistry
» Liver Function
→ Alkaline phosphatase (ALP)
C
► Clinical chemistry
» Metabolic and lipid profile
→ Total cholesterol
D
► Clinical chemistry
» Metabolic and lipid profile
→ Glucose
Female
Male
Sham 3R4F THS(L) THS(M) THS(H) Sham THS(H)

Figure 7. (A) Total protein, (B) alkaline phosphatase (ALP), (C) glucose and (D) total cholesterol levels in serum of in A/J mice following 15 (male mice) or 18 (female mice) months of exposure. *Data are presented as mean±SEM or median±MAD. Lines indicate statistical differences between groups (t-test, p<0.05, not adjusted). Diamonds indicate significant differences between male and female mice (t-test, p<0.05, not adjusted).*

Histopathology and Systems Toxicological Assessment

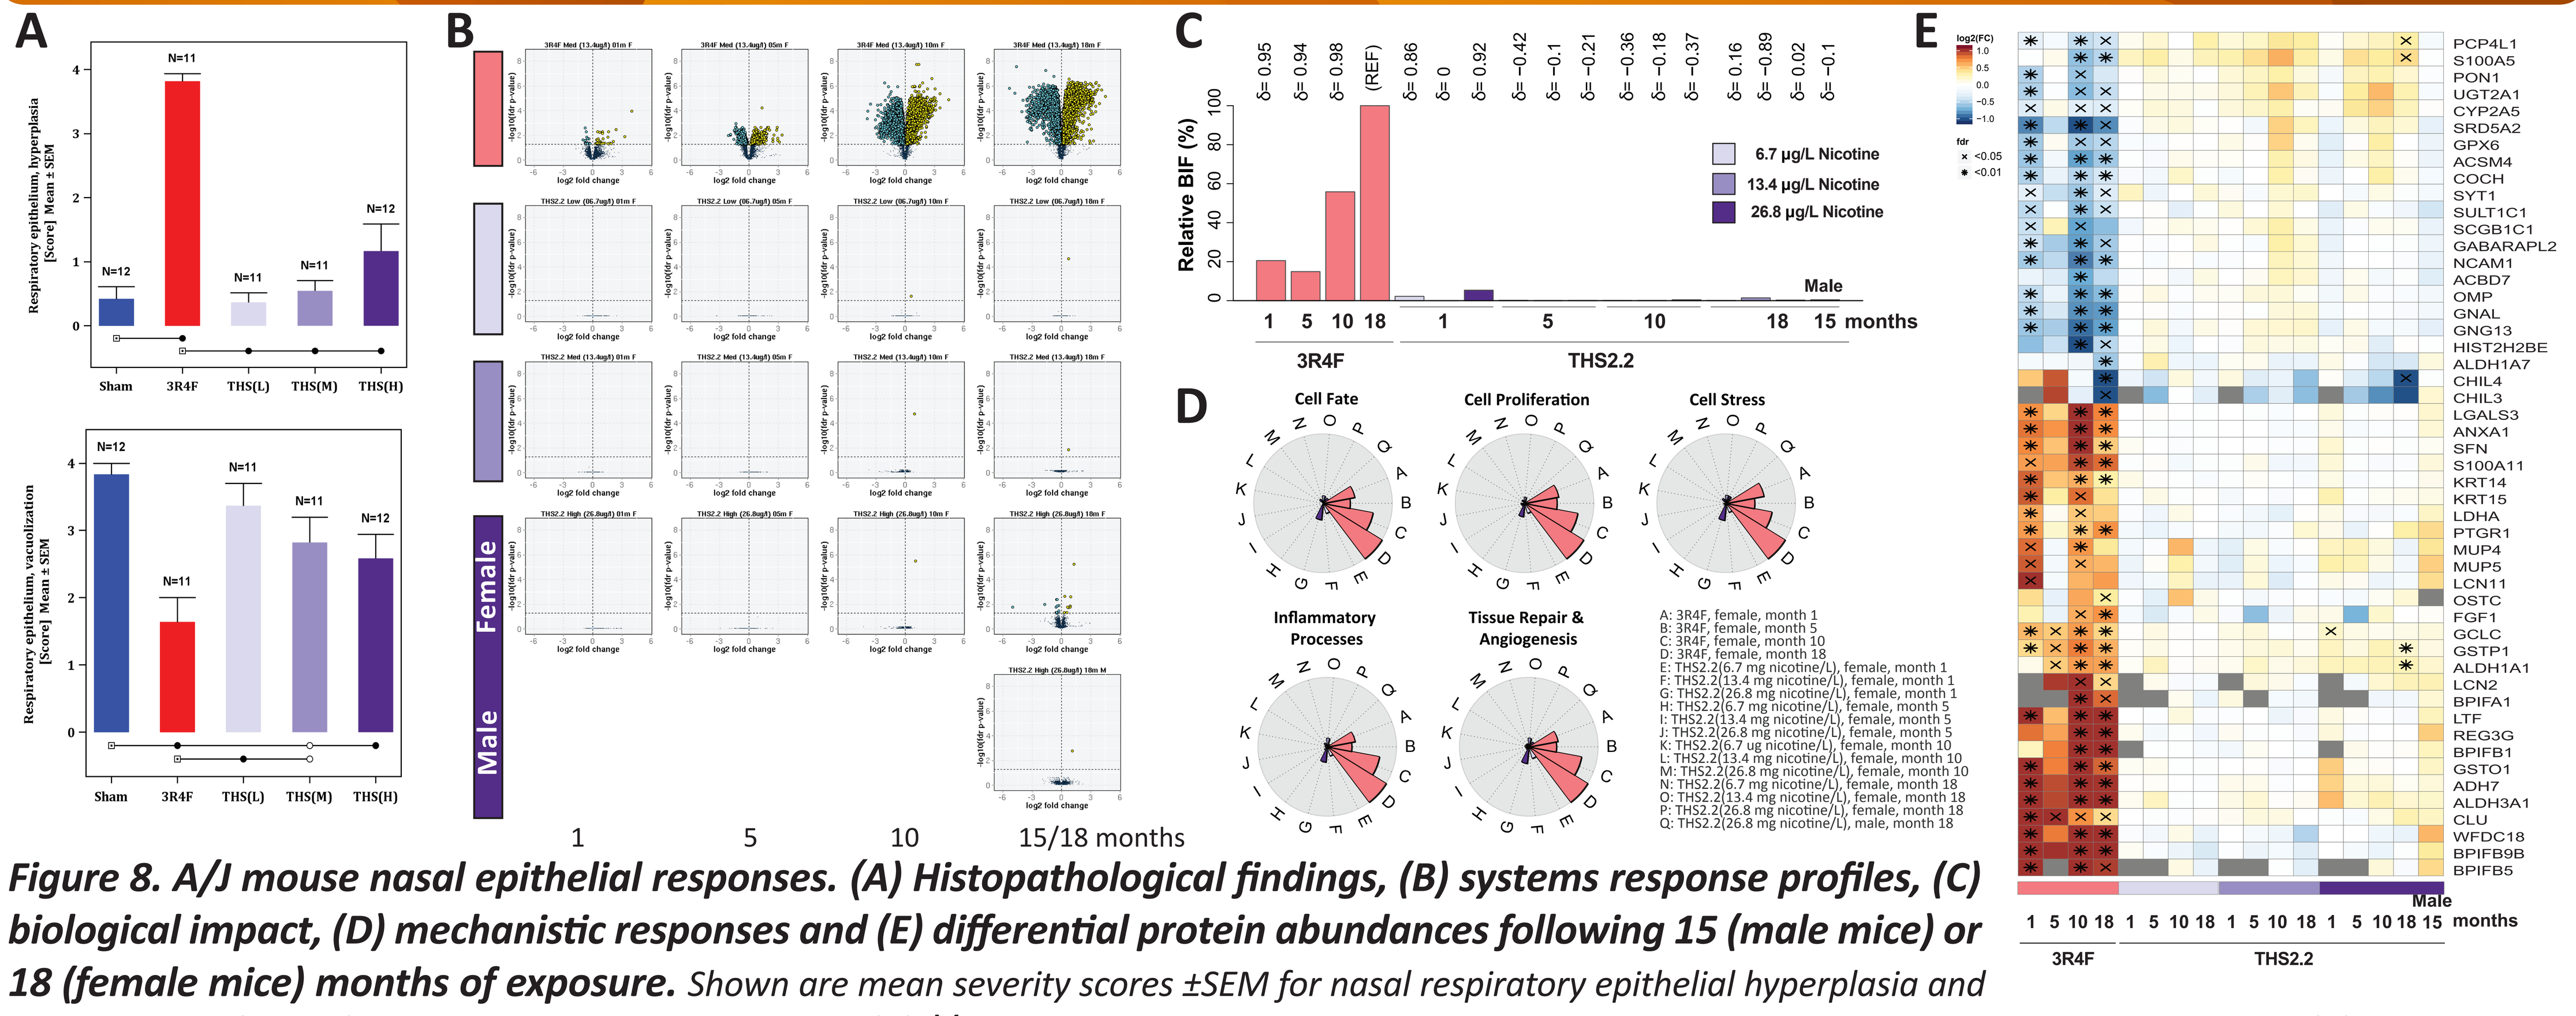
A
B
C
D
E
Female
Male
1 5 10 15/18 months
Relative BIF (%)
6.7 µg/L Nicotine
13.4 µg/L Nicotine
26.8 µg/L Nicotine
3R4F
THS2.2
Cell Fate
Cell Proliferation
Cell Stress
Inflammatory Processes
Tissue Repair & Angiogenesis

Figure 8. A/J mouse nasal epithelial responses. (A) Histopathological findings, (B) systems response profiles, (C) biological impact, (D) mechanistic responses and (E) differential protein abundances following 15 (male mice) or 18 (female mice) months of exposure. *Shown are mean severity scores ±SEM for nasal respiratory epithelial hyperplasia and vacuolization (level 1) of exposed females at month 10 (A). Volcano plots were generated to demonstrate systems responses in the nasal epithelium (B). Each dot in a plot represents a gene. Gene expression changes (x-axis) are expressed as [$\log_2$(fold-change)]; increased when >0 and decreased when <0 as compared with Sham. The statistical significance (y-axis) is represented as [$-\log_{10}$(adj.p-value FDR)]. The horizontal line represents statistical significance [$-\log_{10}(0.05)$]. The bar plot (C) depicts BIF values relative to the maximum response (RBIF = 100%, REF). The δ values (−1 to 1) indicate how similar the underlying network perturbations are with respect to the REF. Scores were computed using transcriptomics profiling data from nasal epithelia. (D) Starplots illustrating the response of the mechanisms, for each the treatment groups (the slices). The surface area of each slice is proportional to the contribution of that mechnism to the overall RBIF of the treatment. The heatmap (E) depicts fold changes in protein abundances (protein symbols listed on the right) relative to corresponding air-exposed controls expressed as log2[fold change (value; fc)]. Increased and decreased fold changes for the top 50 proteins are represented by orange and blue shades, respectively, with darker colors indicating greater absolute fold changes.*

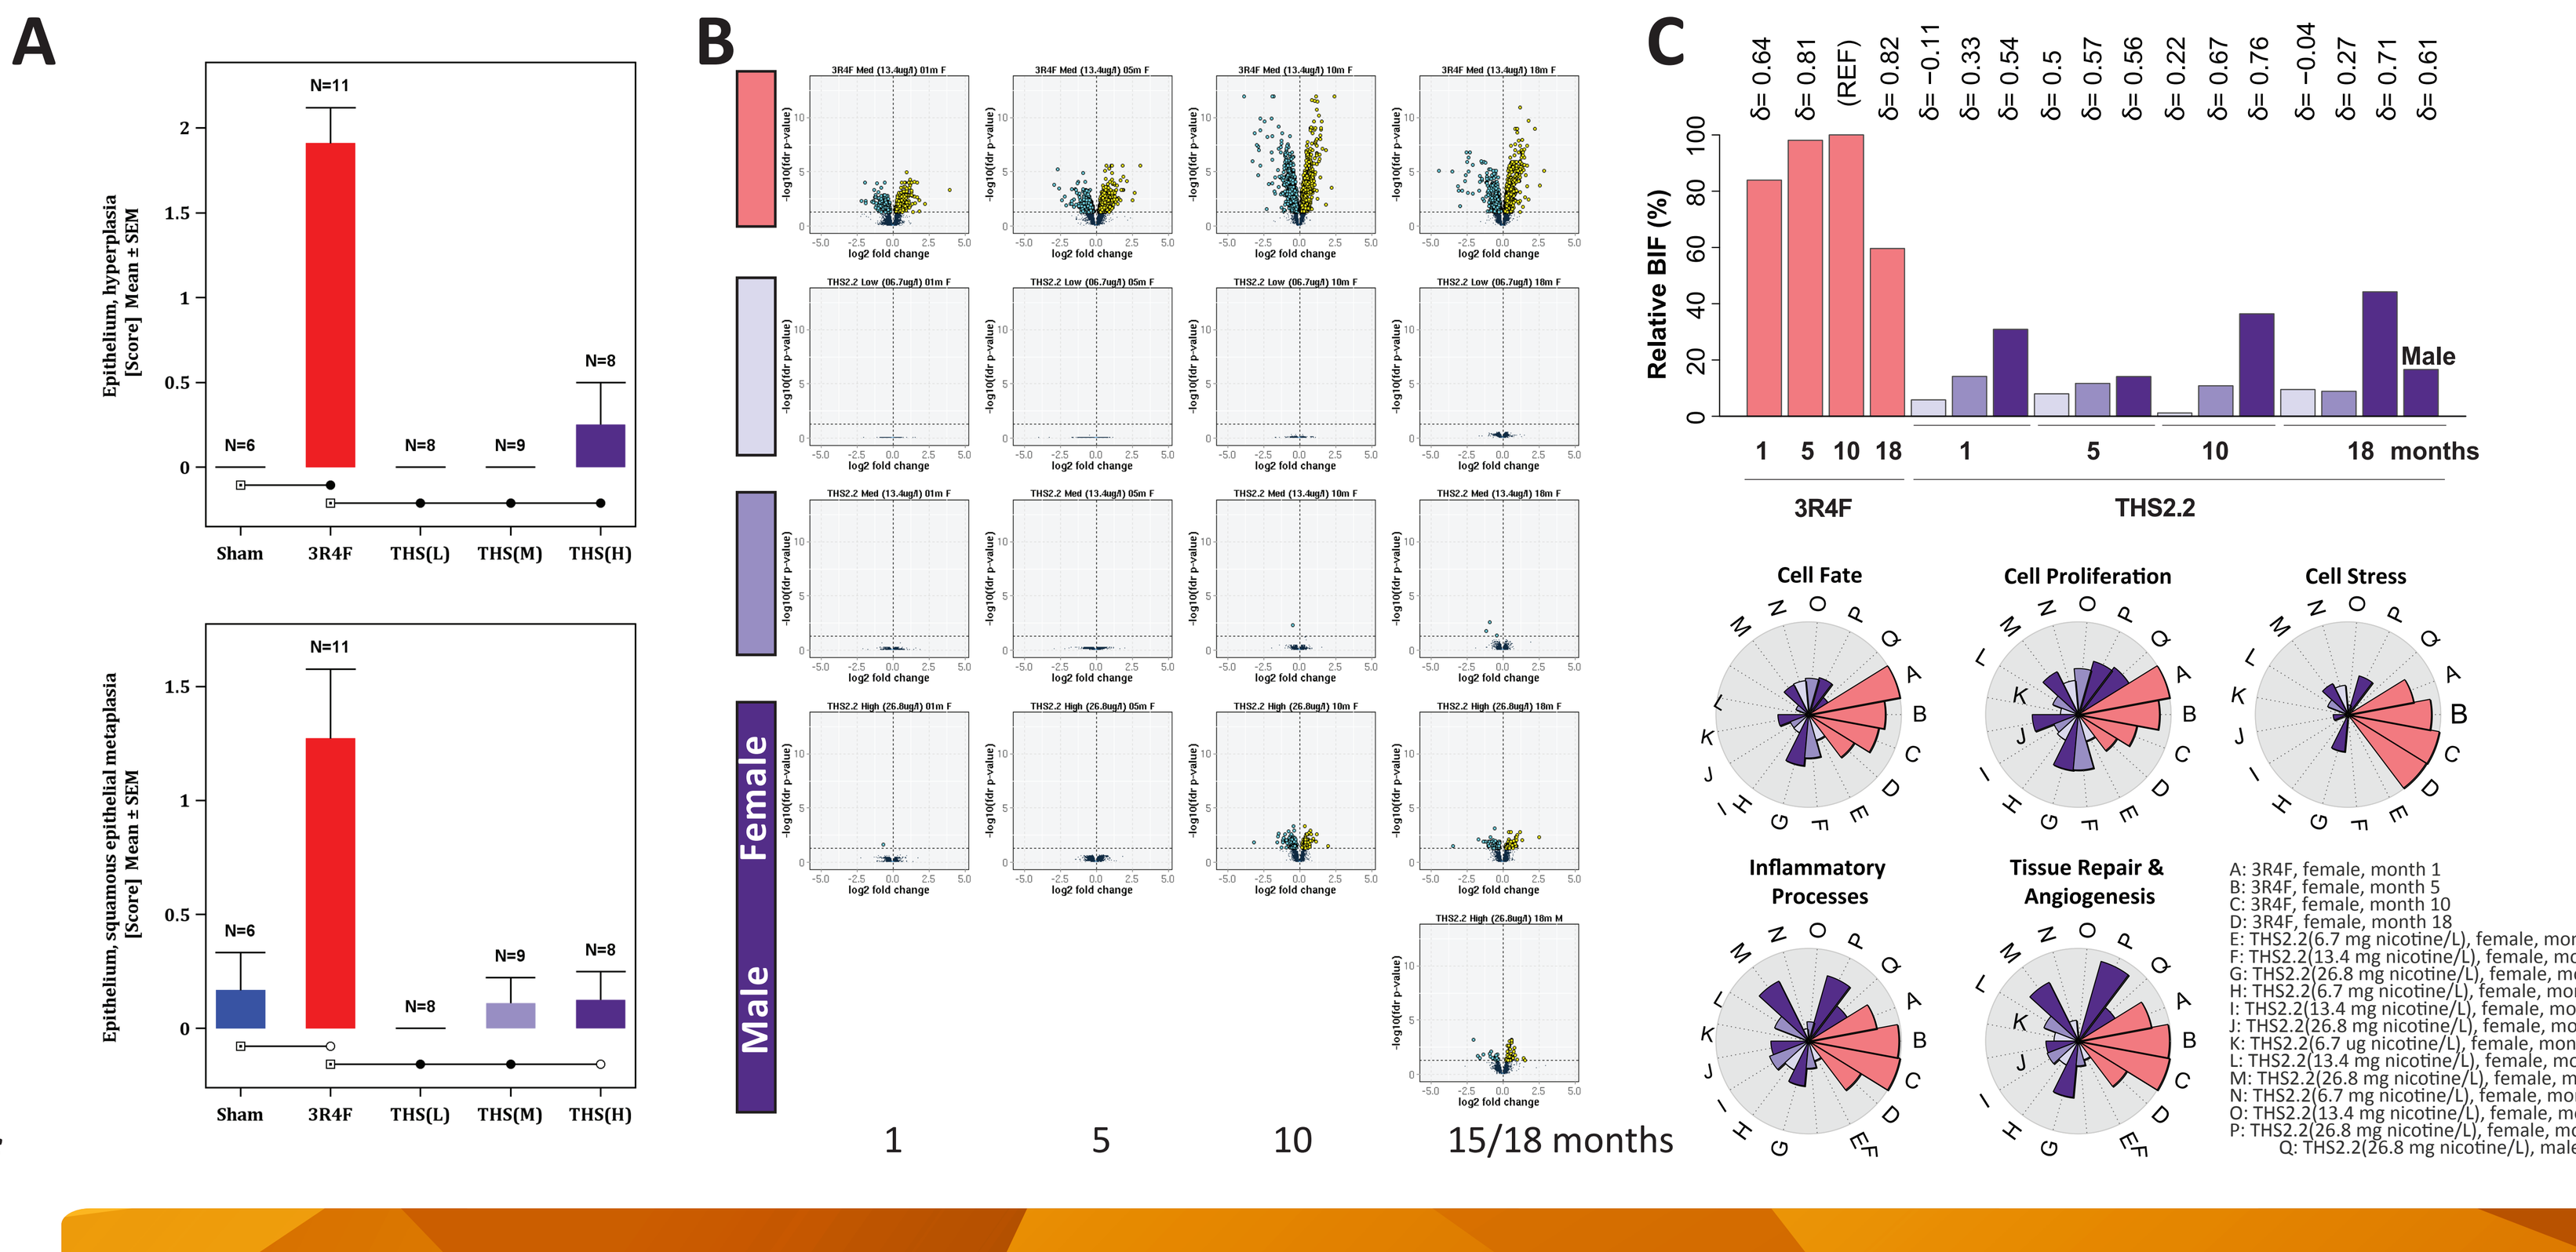
A
B
C
D
Female
Male
1 5 10 15/18 months
Relative BIF (%)
3R4F
THS2.2
Cell Fate
Cell Proliferation
Cell Stress
Inflammatory Processes
Tissue Repair & Angiogenesis

Figure 9. A/J mouse tracheal epithelial responses. (A) Histopathological findings at month 10, (B) systems response profiles, (C) biological impact and (D) mechanistic response following 15 (male mice) or 18 (female mice) months of exposure.

Conclusions

The characterization of the aerosol components as well as biomarkers of aerosol uptake indicated that the delivery and uptake of 3R4F CS and THS2.2 aerosol by the mice were in line with expectations. The test atmosphere was well tolerated as reflected by the progressive weight gain over time. Exposure to CS induced changes in red and white blood cell profiles, liver function and metabolic parameters, but only minimal changes were observed following THS2.2 aerosol exposure. Exposure to CS also resulted in histopathological changes in the nose and trachea, while no significant effects were observed following THS2.2 aerosol exposure. Significant changes in the nasal and tracheal epithelial transcriptomes and the nasal epithelial proteome were observed following CS exposure. The perturbed mechanisms are consistent with CS exposure, but only minor perturbations were noted in nose and trachea following THS2.2 aerosol exposure. With the histopathological evaluation of the lungs and other organs still ongoing, no final conclusion regarding the chronic toxicity and carcinogenicity of THS2.2 can be drawn at this point.

PMI SCIENCE
PHILIP MORRIS INTERNATIONAL
www.pmiscience.com

EUROTOX 2017
10-13 September 2017
Bratislava, Slovakia

Competing Financial Interests
The research described in this poster was sponsored by Philip Morris International