Cilia of human MucilAir™ are less impacted by the exposure to the aerosol of a candidate modified risk tobacco product than to whole smoke from conventional cigarettes *in vitro*

Stefan Frentzel, Davide Grandolfo, Diana Kuehn, Shoaib Majeed, Patrice Leroy, Manuel C Peitsch, and Julia Hoeng
Philip Morris International R&D, Philip Morris Products S.A., Neuchâtel, Switzerland (part of Philip Morris International group of companies)

#P283

Abstract

Mucociliary clearance is an important defense mechanism that mediates removal of foreign particles and chemicals from the airways. Cilia beating thereby plays a key role that determines the rate of mucus clearance and thus constitutes a vital function of respiratory epithelia. Cigarette smoke has been reported to adversely impact cilia function *in vitro* and *in vivo*, by changing cilia beating frequency (CBF) or impairing ciliogenesis. To monitor CBF, semi-automated methods such as CiliaFA combine high-speed video recording with the ability to determine CBF. Using CiliaFA, we were able to confirm that the MucilAir™ CBF can be modulated in vitro by either 100 µM isoproterenol or a temperature shift to 4°C. Moreover, *in vitro* exposure of MucilAir™ with whole smoke from conventional cigarettes (3R4F at 0 mg/L, 0.15 mg/L and 0.25 mg/L nicotine) caused a decrease in the total surface area of the culture showing active cilia beating. Cilia on the epithelial cell surface that were detected to be still active after 3R4F exposure, showed variable beat frequencies, ranging from normal to decreased CBF. Compared to 3R4F cigarette smoke exposure, the effect of equivalent concentrations (based on nicotine) of a candidate modified risk tobacco product (MRTP: THS2.2) aerosol was much less pronounced, i.e. the total surface area of cilia beating as well as beating frequency was less impacted by THS2.2. Overall, this study clearly discriminated the effects of THS2.2 from the deleterious impact of 3R4F on cilia beating.

Introduction

Synchronized beating of cilia on epithelial cells of human airways mediates transport of inhaled pathogens and potential noxious particles trapped in the mucus layer (Figure 1) towards the pharynx, where they eventually become swallowed or expectorated [1]. Effective cilia beating is therefore a key requirement for functional mucociliary clearance in the lower (broncho-tracheal) and upper respiratory tract (laryngeal - nasal). Defects in cilia function as a consequence of disorders such as primary ciliary dyskinesia can lead to persistent respiratory infection [2, 3]. In addition, exposure to cigarette smoke can impair mucociliary transport by inducing mucus hypersecretion and goblet cell hyperplasia and thus worsen airflow obstruction and predisposes to bacterial infection [4]. Several studies showed that exposure of the ciliated epithelium to particles of cigarette smoke results in a significant decrease in CBF [5, 6]. Although other studies did not find any difference in ciliary beating frequency between smokers and nonsmokers, they confirmed that mucociliary transport was disrupted in regular smokers [7]. In the following study we investigated the effect of cigarette smoke on CBF *in vitro*, measured upon expose of human nasal MucilAir™. Also, this study aimed to evaluate the impact of a MRTP aerosol on CBF. As not only beating frequency is an informative parameter to judge on the mucociliary clearance capacity, the overall activity of cilia on MucilAir™ was measured too (represented as Cilia power; this parameter was introduced as it is more accurate compared to "% area active" shown in the CiliaFA report). Finally, general cytotoxicity as well as changes in the tissue morphology were assessed.

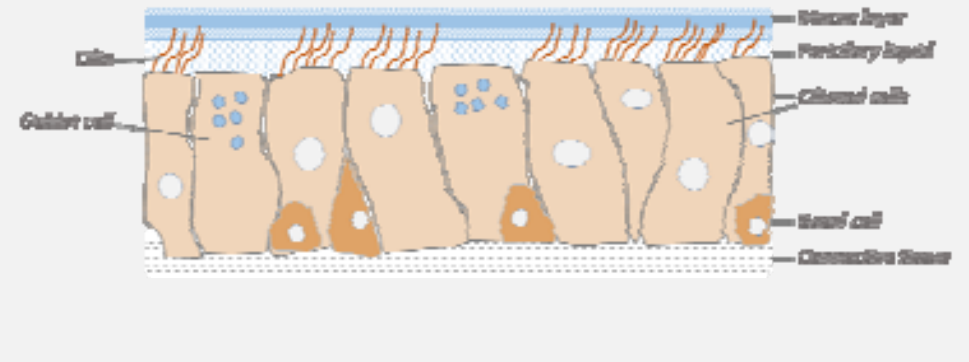

Figure 1) Schematic representation of cellular and non-cellular components constituting the mucociliary clearance system in human airways.

Materials & Methods

Human nasal MucilAir™ 3D-organotypic tissue cultures (without fibroblasts) used in this study were purchased from Epithelix Sàrl (Geneva, Switzerland). Prior to the delivery date, tissue inserts were cultured at the air-liquid interface for 39 consecutive days and fully differentiated into pseudostratified epithelia. Donor characteristics: 53 years old, male, non-disease, non-smoker, Caucasian. At the day of arrival, an apical wash followed by Transepithelial Electrical Resistance (TEER) measurement has been carried out, to remove excessive mucus and to check for tissue integrity (TEER ≥ 200 Ω/cm²). For aerosol exposure, MucilAir™ where inserted into a Vitrocell™ 24/48 exposure system (Vitrocell Systems GmbH, Waldkirch, Germany) and exposed for 28 minutes, at 37°C, 60% relative humidity (Health Canada intense, puffing protocol). Aerosols were generated with a SM2000 smoking machine (Philip Morris International). Reference smoke was generated from 3R4F reference cigarettes (University of Kentucky, www.ca.uky.edu/refcig) using a 30-port carousel smoking machine type SM2000 (SM2000; Philip Morris, International) with a Programmable Dual Syringe Pump (PDSP) connected to the Vitrocell™ 24/48. Another 30-port carousel smoking machine type SM2000 with a PDSP was used to generate the test aerosol from a candidate MRTP.

Materials & Methods cont.

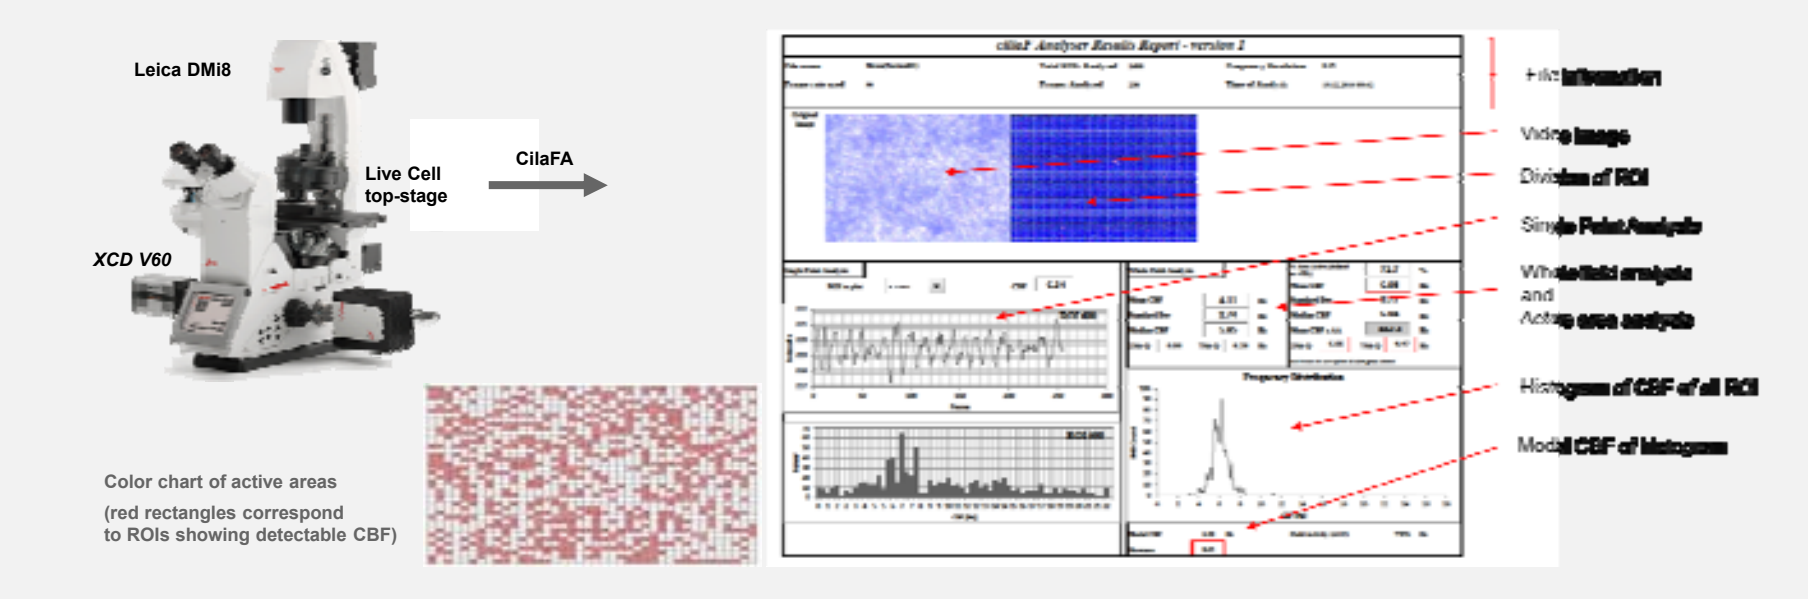
Leica DMi8
Live Cell top-stage
CilaFA
XCD V60
Color chart of active areas
(red rectangles correspond to ROIs showing detectable CBF)

Figure 2) Setup for measuring CBFon MucilAir™ using CiliaFA (report sheet depicted right side)

To determine CBF, human nasal MucilAir™ were placed in a top-stage incubation chamber at controlled temperature (37°C; Life Cell Instruments, Seoul, Korea) and allowed to equilibrate for 5 minutes before reading. Beating cilia were recorded using a Sony XCD V60 digital high-speed video camera connected to an inverted microscope system (Leica DMi8), at a rate of 90 frames per second using a × 4 objective. A total of 512 frames were recorded from at least two visual fields (insert center and peripheral). To analyse beating frequencies the freely available automated software CiliaFA was used, which runs on the open-source software (ImageJ) coupled to Microsoft Excel. It extracts pixel intensities of region of interests (ROIs) over time from the AVI file previously recorded, which is then further used for Fast Fourier transformation (FFT) in Excel to infer CBF between 3 and 52 Hz. The resulting report sheet shows the CBF values for each individual ROI or the image as a whole. To determine more accurate CBF by CiliaFA outputs, the following methods have been applied: All1600 spectra (FFT magnitudes) related to the 1600 ROI defined in the CiliaF Analyser Results Report were aggregated (summed) to obtain a single spectrum. Spectra from different visual fields of the same insert were averaged. For a noise reduction, the spectra were smoothened by using a moving average on 10 points (the delta between points is around 0.18Hz). CBF was then determined according to the highest FFT magnitude in the spectra. A cut-off was set to ≥ 2.5 Hz, in order to avoid large peaks for extremely low frequencies. Also, if the highest frequency of the FFT magnitude was less than 1.25*the lowest magnitude in the spectrum (the noise) then this value was omitted. The FFT power normalized to ambient noise is then calculated as: (FFT magnitude - lowest FFT magnitude)^2 / lowest FFT magnitude. Next, the FFT power was integrated/summed over all the frequencies (again only for frequencies above 2.5Hz). The summed FFT power normalized to ambient noise was further normalized to sham (=100%) for each exposure run independently (n=9, except for: 3R4F 0.25 mg/L - pre-exposure n=3; THS2.2 0.15 mg/L – 0h post-exposure n=8; 3R4F 0.25 mg/L - 24h post-exposure n=6; THS2.2 0.15 mg/L – 24h post-exposure n=8; 3R4F 0.25 mg/L – 48h post-exposure n=5; THS2.2 0.27 mg/L – 48h post-exposure n=8). For Statistical comparisons: Paired TTEST were used to compare two doses of the same product (paired by smoke run) and TTEST adjusting for heterogeneous variance (Satterthwaite correction) were used to compare THS2.2 to 3R4F.

Results

Modulation of CBF using known controls

a)

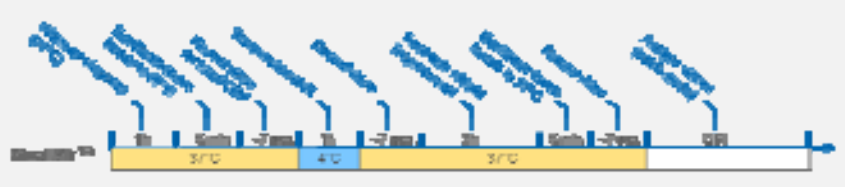

b)

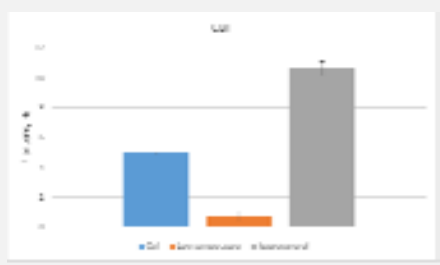

c)

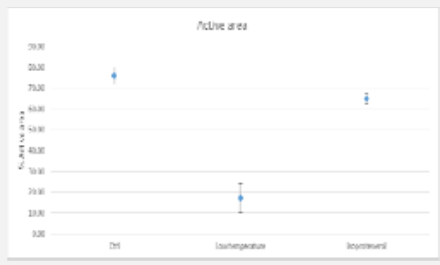
Active area

Figure 3) Nasal MucilAir™ incubated with positve control stimulant (isoproterenol) or decreased temperature

a) Experimental design/workflow. b) Exposure to isoproterenol (100µM) applied to the basolateral side of MucilAIr™ for 3 hours produced an increase in CBF of approximately 5 Hz in 3D culture of nasal epithelial cells, while incubation at 4°C for 30 minutes produced a decrease in CBF of approximately 4 Hz. c) The decrease in CBF correlated with a decrease in the total area with active cilia beating.

Effect of aerosol exposure on CBF (I)

a)

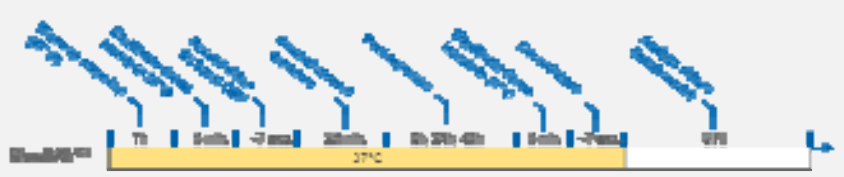

b)

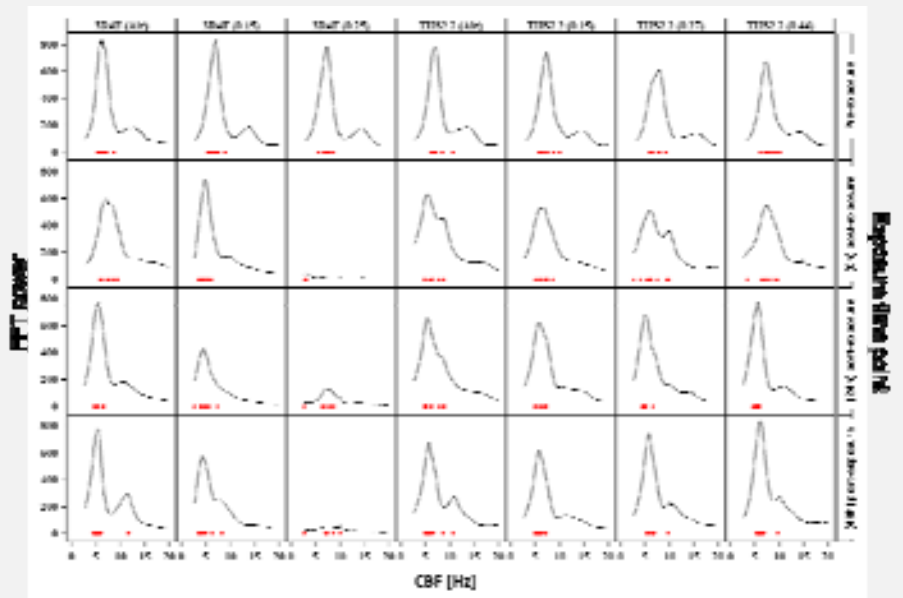
FFT power
CBF [Hz]
Exposure time point

Figure 4) CBF distributions upon nasal MucilAir™ exposure to different doses of 3R4F whole cigarette smoke or THS2.2 MRTP aerosol

a) Experimental design/workflow. b) Diagrams show the distribution of determined CBF's (X-axis) aggregated from all exposure runs of individual test aerosols. The topline indicates the test aerosol to which MucilAir™ was exposed to and its respective dose, given as mg/L nicotine present in the applied aerosol (3R4F: reference cigarette smoke, THS2.2 aerosol). FFT power determined for cilia beating frequencies can be seen from the Y-axis. Diagrams from top to down belong to CBF distributions determined at different exposure time points (indicated right). Red dots represent frequencies of highest FFT magnitude determined for different runs.

Results – cont.

Effect of aerosol exposure on CBF (II) and cytotoxicity

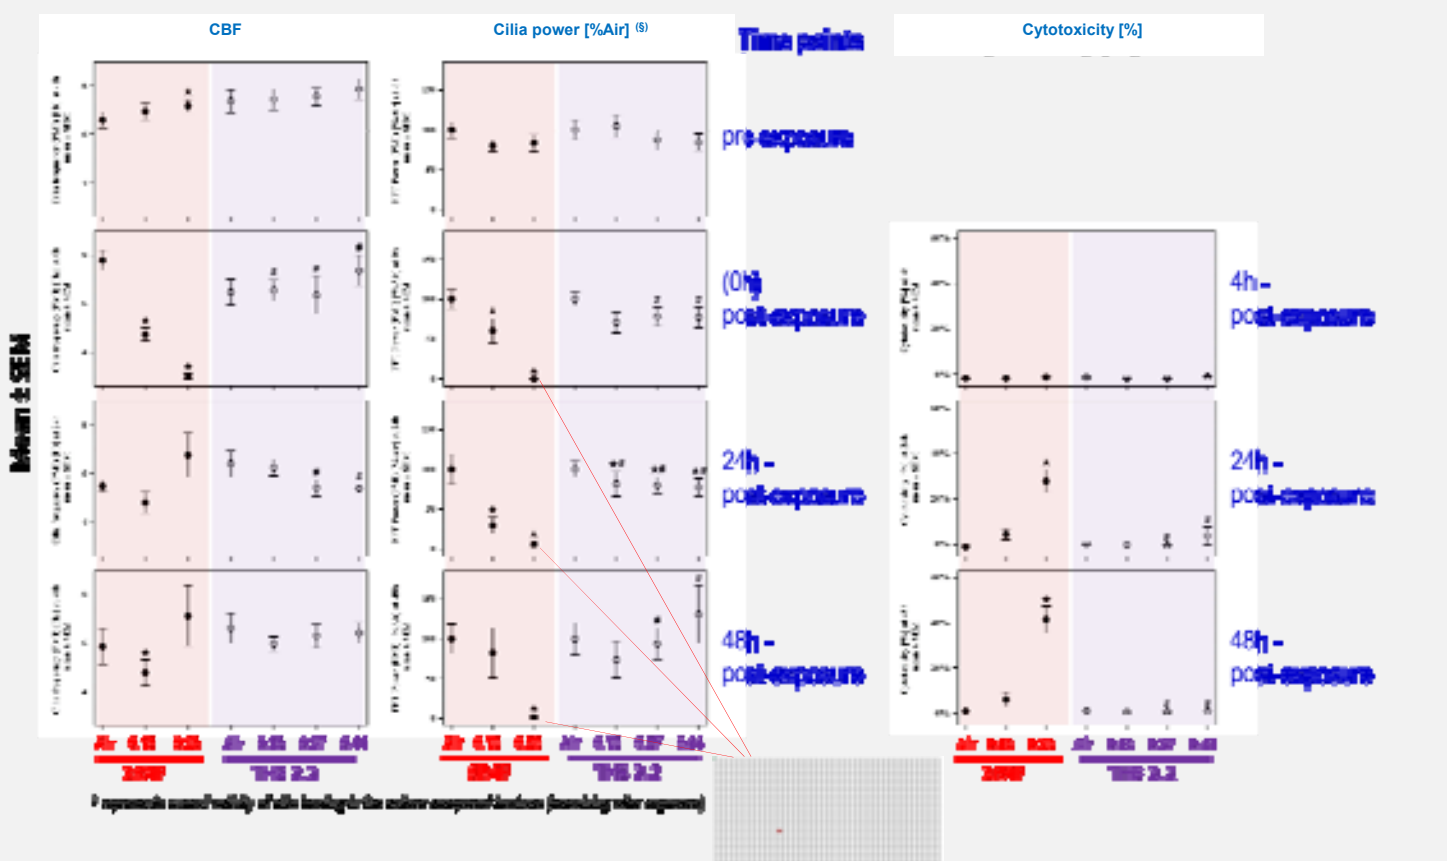
CBF
Cilia power [%Air]
Time points
Cytotoxicity [%]
Mean ± SEM
pre-exposure
(0h) post-exposure
4h - post-exposure
24h - post-exposure
48h - post-exposure
3R4F
THS 2.2

Figure 5) Effect of different test aerosols on CBF (left graphs), Cilia power (middle graphs) and resulting cytotoxicity (right graphs) upon exposure of nasal MucilAir™ to 3R4F and THS2.2 aerosol.

X-axis indicate applied doses of respective aerosols tested (3R4F reference cigarette smoke or aerosol from THS2.2 MRTP), given as mg/L nicotine present in the applied aerosol. Y-axis correspond to mean values ± SEM of CBF, Cilia power [%] or Cytotoxicity [%] respectively. A star indicates a difference versus corresponding sham, a hash a difference between: - THS 0.15 mg/L and 3R4F 0.15 mg/L, - THS 0.27 mg/L and 3R4F 0.25 mg/L or - THS 0.44 mg/L and 3R4F 0.25 mg/L (p≤0.05). Cytotoxicity was determined by measuring the release of adenylate kinase (AK) into the basolateral medium, using the ToxiLight™ Non-destructive Cytotoxicity BioAssay Kit (Lonza, Basel, Switzerland). The insert at the lower end of the figure shows a color chart from the CiliaFA report, which is typical for a high dose 3R4F exposure, with almost no ROI left in the visual field showing detectable CBF (single red rectangle).

Effect of aerosol exposure on tissue morphology

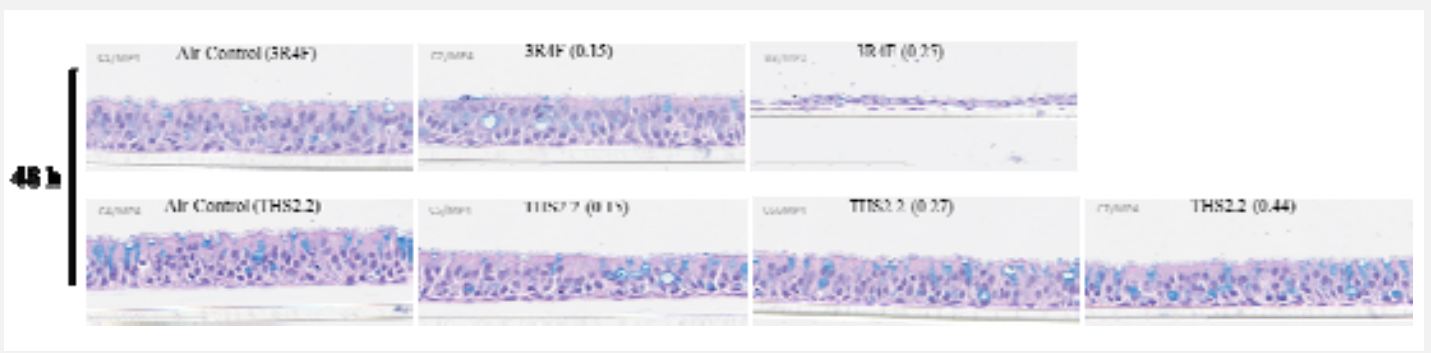
Air Control (3R4F)
3R4F (0.15)
Air Control (THS2.2)
THS2.2 (0.44)
48 h

Figure 6) Histological analysis (combined H&E - Alcian blue staining) of nasal MucilAir™ after exposure to different doses of 3R4F cigarette smoke and THS2.2 aerosol (48h post-exposure)

Upper pictures = exposure to 3R4F cigarette smoke; lower pictures = exposure to THS2.2 aerosol. Doses (mg/L nicotine) are in brackets.

Summary

- Cigarette smoke from 3R4F shows a clear dose-dependent effect on both, cilia beating frequency and Cilia power after *in vitro* exposure of MucilAir™ cultures
- The effect on CBF seems to be partially reversible for the low 3R4F dose, at 24-48h post-exposure
- CBF at high 3R4F dose appears to recover, however at this dose Cilia power on MucilAir™ was reduced to almost 0% and coincided with high cytotoxicity and large tissue damage
- Compared to the matching doses of 3R4F cigarette smoke, exposure to aerosol from THS2.2 shows only small effects, if any, on CBF and Cilia power

References

1. Fokkens, W. J., et al. (2000). Paediatr. Respir. Rev. 1:336-341
2. Noone P.G., et al. (2004). Am J Respir Crit Care Med. 169:459-67
3. Verkman A.S., et al. (2003). Am. J. Physiol. Cell Physiol. 284:2–15
4. Mebratu, Y.A., et al. (2011). Am J Respir Crit Care Med. 183(11): p. 1531-8
5. Top E.A.V., et al. (2005).Clin Exp Res. 29:882-7
6. Cohen N.A., et al. (2009). Laryngoscope. 119:2269-74
7. Stanley P.J., et al. (1986). Thorax. 41:519-23

PMI RESEARCH & DEVELOPMENT

Society of Toxicology
55th Annual Meeting and ToxExpo
March 13 – 17, 2016, New Orleans

Philip Morris Products S.A., Neuchâtel, Switzerland (part of Philip Morris International group of companies)