GENERATION OF **REPORTER GENE MODELS** BASED ON GENETICALLY ENGINEERED PROGENITOR CELLS USING CRISPR–Cas9 TECHNOLOGY

Marion Alriquet, David Bovard, Remi Dulize, David Bornand, James Battey, Emmanuel Guedj, Nicolas Sierro, Claudius Pak, Shoaib Majeed, Karsta Luettich, Marco van der Toorn, Stefan Frentzel, Julia Hoeng

PMI R&D, Philip Morris Products S.A., Quai Jeanrenaud 5, CH-2000 Neuchâtel, Switzerland

Introduction and objectives

Three-dimensional (3D) bronchial organotypic models

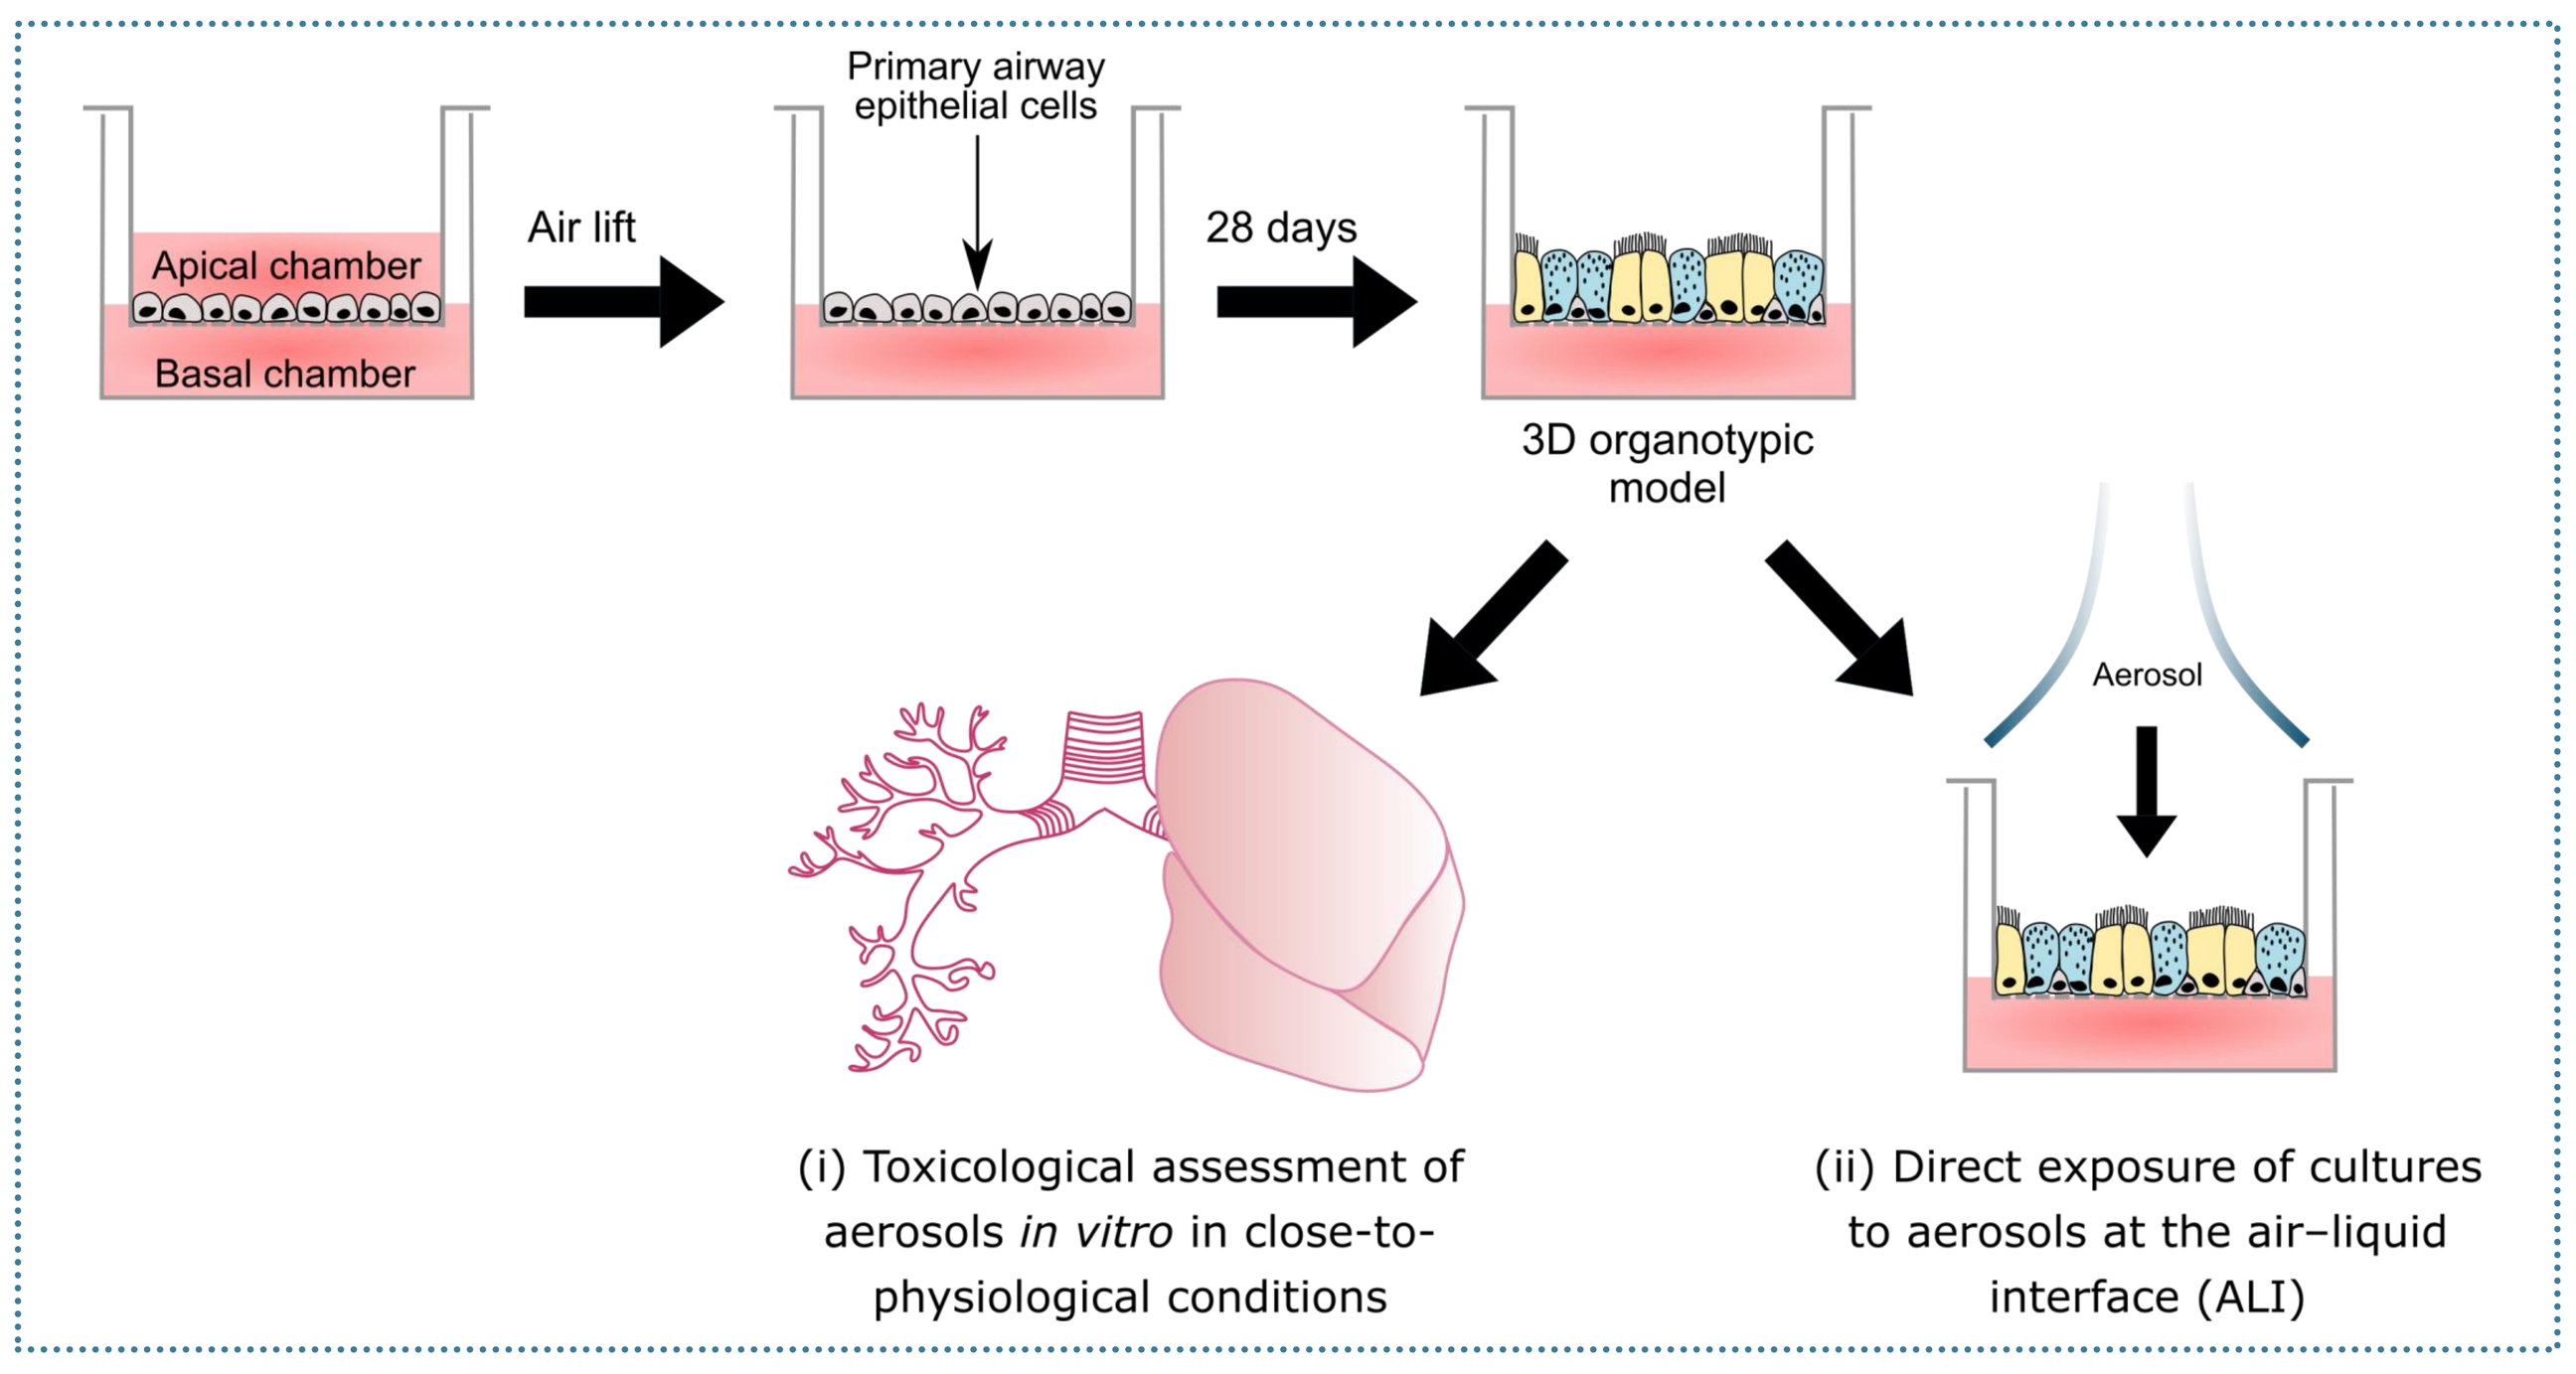
Primary airway epithelial cells
Apical chamber
Basal chamber
Air lift
28 days
3D organotypic model
Aerosol
(i) Toxicological assessment of aerosols *in vitro* in close-to-physiological conditions
(ii) Direct exposure of cultures to aerosols at the air–liquid interface (ALI)

Figure 1. Schematic representation of a 3D model derived from primary bronchial epithelial cells. Basal cells are seeded on the porous membrane of an insert. The apical medium is removed (air lift). At 28 days after air lift, the tissues are fully differentiated and ready to use for toxicology studies.

Objective

Generate **reporter gene models** using primary airway cells differentiated into 3D ALI cultures
- Monitor the effect of aerosol exposure on stress response genes in a **non-invasive manner**
- Use **CRISPR–Cas9** to tag the **oxidative stress** marker HMOX1 with a fluorescent protein
- Test the functionality of the CRISPR–Cas9 reporter constructs in **a simple cell system**: HEK293

Results

- One HEK293 **reporter cell line** characterized; HMOX1–GFP inducibility upon oxidative stress induction demonstrated in this cell line

Next step

- Application of the methodology **to primary airway cells** to develop 3D reporter models

Genome engineering strategy

- Several gRNAs were designed using CHOPCHOP (Labun et al., 2019) and screened in order to increase the chance of success of the genome engineering (Fig. 2).
- The genome engineering strategy is described in Figure 3.

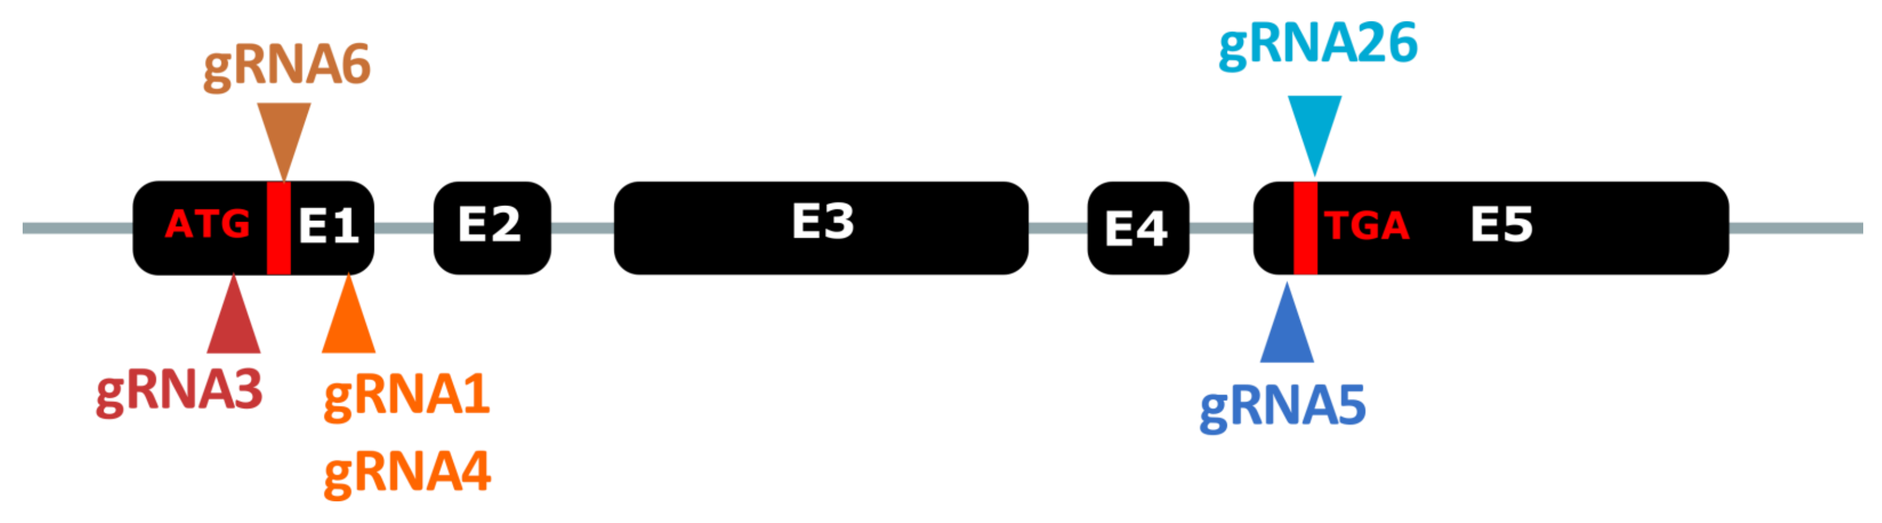
gRNA6
gRNA26
ATG
E1
E2
E3
E4
TGA
E5
gRNA3
gRNA1
gRNA4
gRNA5

Figure 2. Schematic representation of the HMOX1 gene. The cutting sites of the 6 gRNAs that were tested are indicated. gRNA, guide RNA; ATG, start codon; TGA, stop codon; En, exon number n

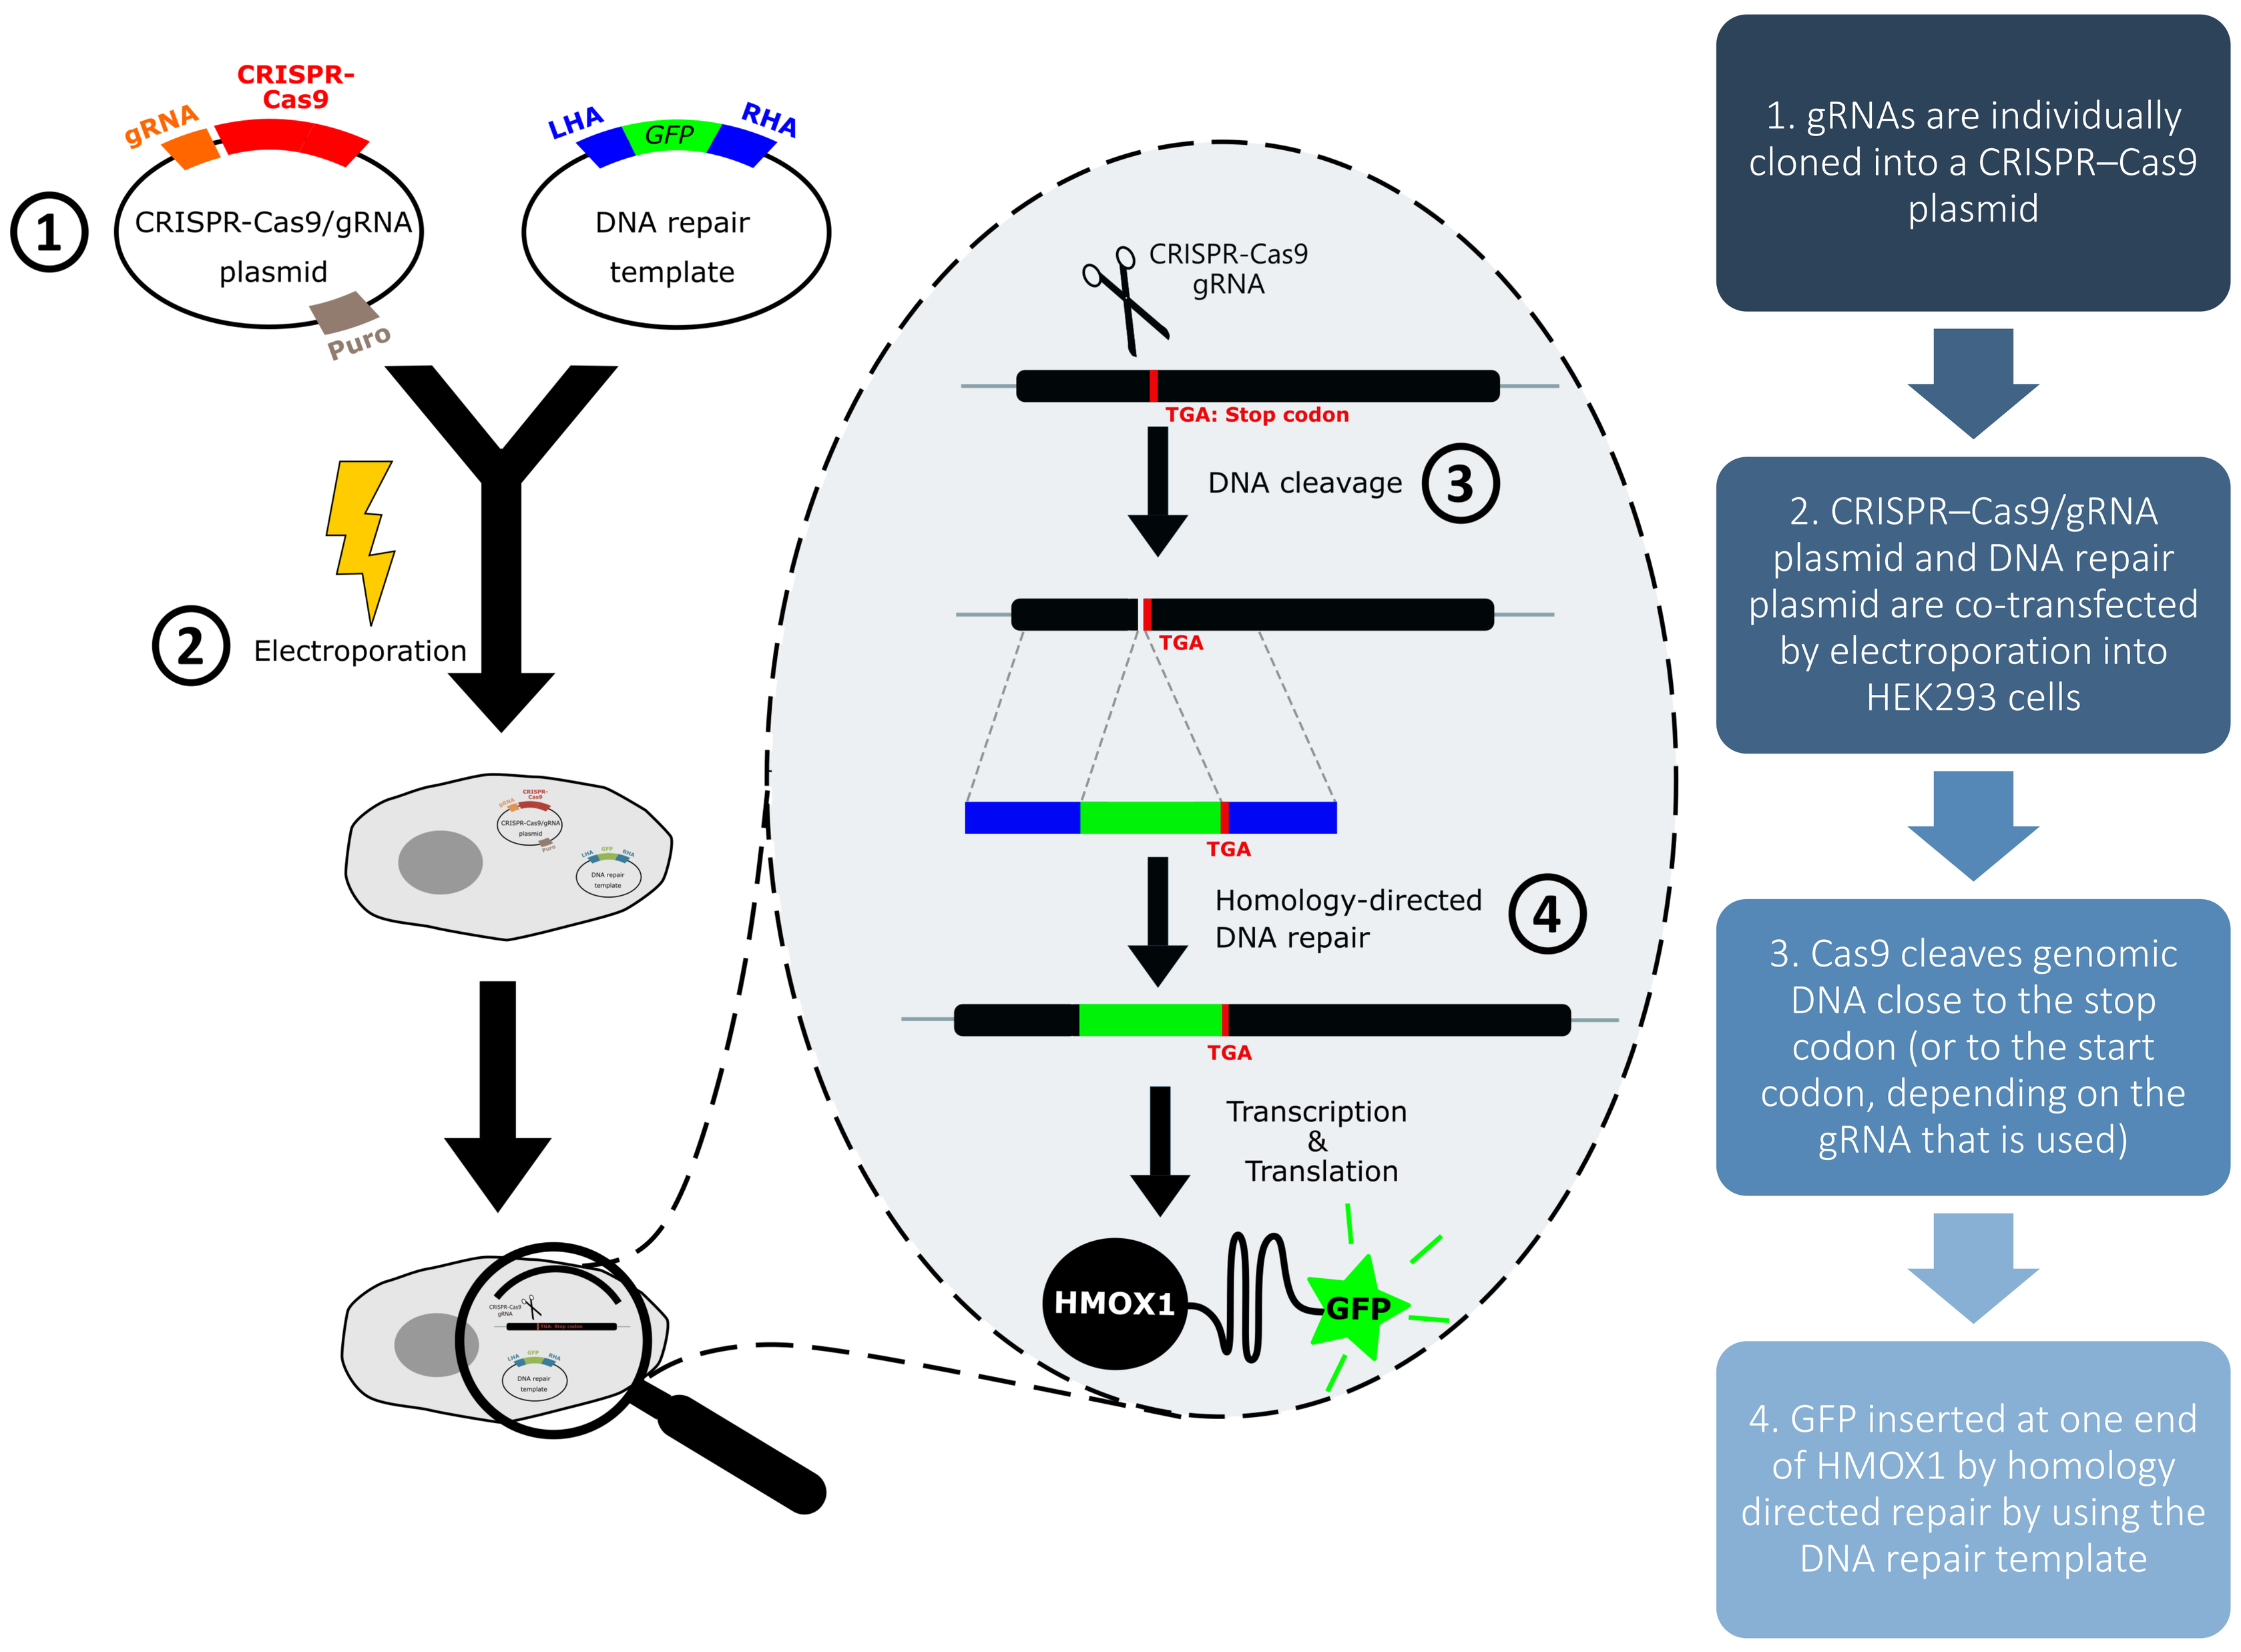
CRISPR-Cas9
gRNA
CRISPR-Cas9/gRNA plasmid
Puro
LHA
GFP
RHA
DNA repair template
1
2
Electroporation
CRISPR-Cas9 gRNA
TGA: Stop codon
DNA cleavage
3
TGA
Homology-directed DNA repair
4
Transcription & Translation
HMOX1
GFP
1. gRNAs are individually cloned into a CRISPR–Cas9 plasmid
2. CRISPR–Cas9/gRNA plasmid and DNA repair plasmid are co-transfected by electroporation into HEK293 cells
3. Cas9 cleaves genomic DNA close to the stop codon (or to the start codon, depending on the gRNA that is used)
4. GFP inserted at one end of HMOX1 by homology directed repair by using the DNA repair template

Figure 3. Schematic view of the genome engineering strategy applied to HEK293 cells.
gRNA, guide RNA; Puro, puromycin resistance gene; LHA, left homology arm; RHA, right homology arm; GFP, green fluorescent protein

Conclusions

- ✓ CRISPR–Cas9 was used to generate **reporter gene models** that will allow convenient *in vitro* aerosol toxicity testing by fluorescence signal measurement.
- ✓ The methodology was evaluated in the immortalized cell line HEK293 with **HMOX1 tagged at the C terminal with GFP**.
- ✓ HMOX1–GFP is induced in a dose-dependent manner upon treatment with the **oxidative stress inducer hemin**.
- ✓ The reporter cell line is responsive to treatment with **cigarette smoke extract**.
 - ↘ Next step: Apply the methodology to **normal human bronchial epithelial (NHBE) cells** by using gRNA26 (the most efficient gRNA in our genome-editing approach).
 - ↘ If gene editing is successful in NHBE cells, 3D models will be generated to measure oxidative stress induction by determining **fluorescence intensity** upon aerosol exposure.

Results

1 **Identification of positive clones:** Genomic DNA of HEK293–HMOX1–GFP clones was extracted and used as a template for control PCR (Fig. 4). One of the positive clones that expressed HMOX1 tagged at the C-terminal end with GFP (HMOX1–GFP) was used for subsequent experiments. This clone was generated by using gRNA26.

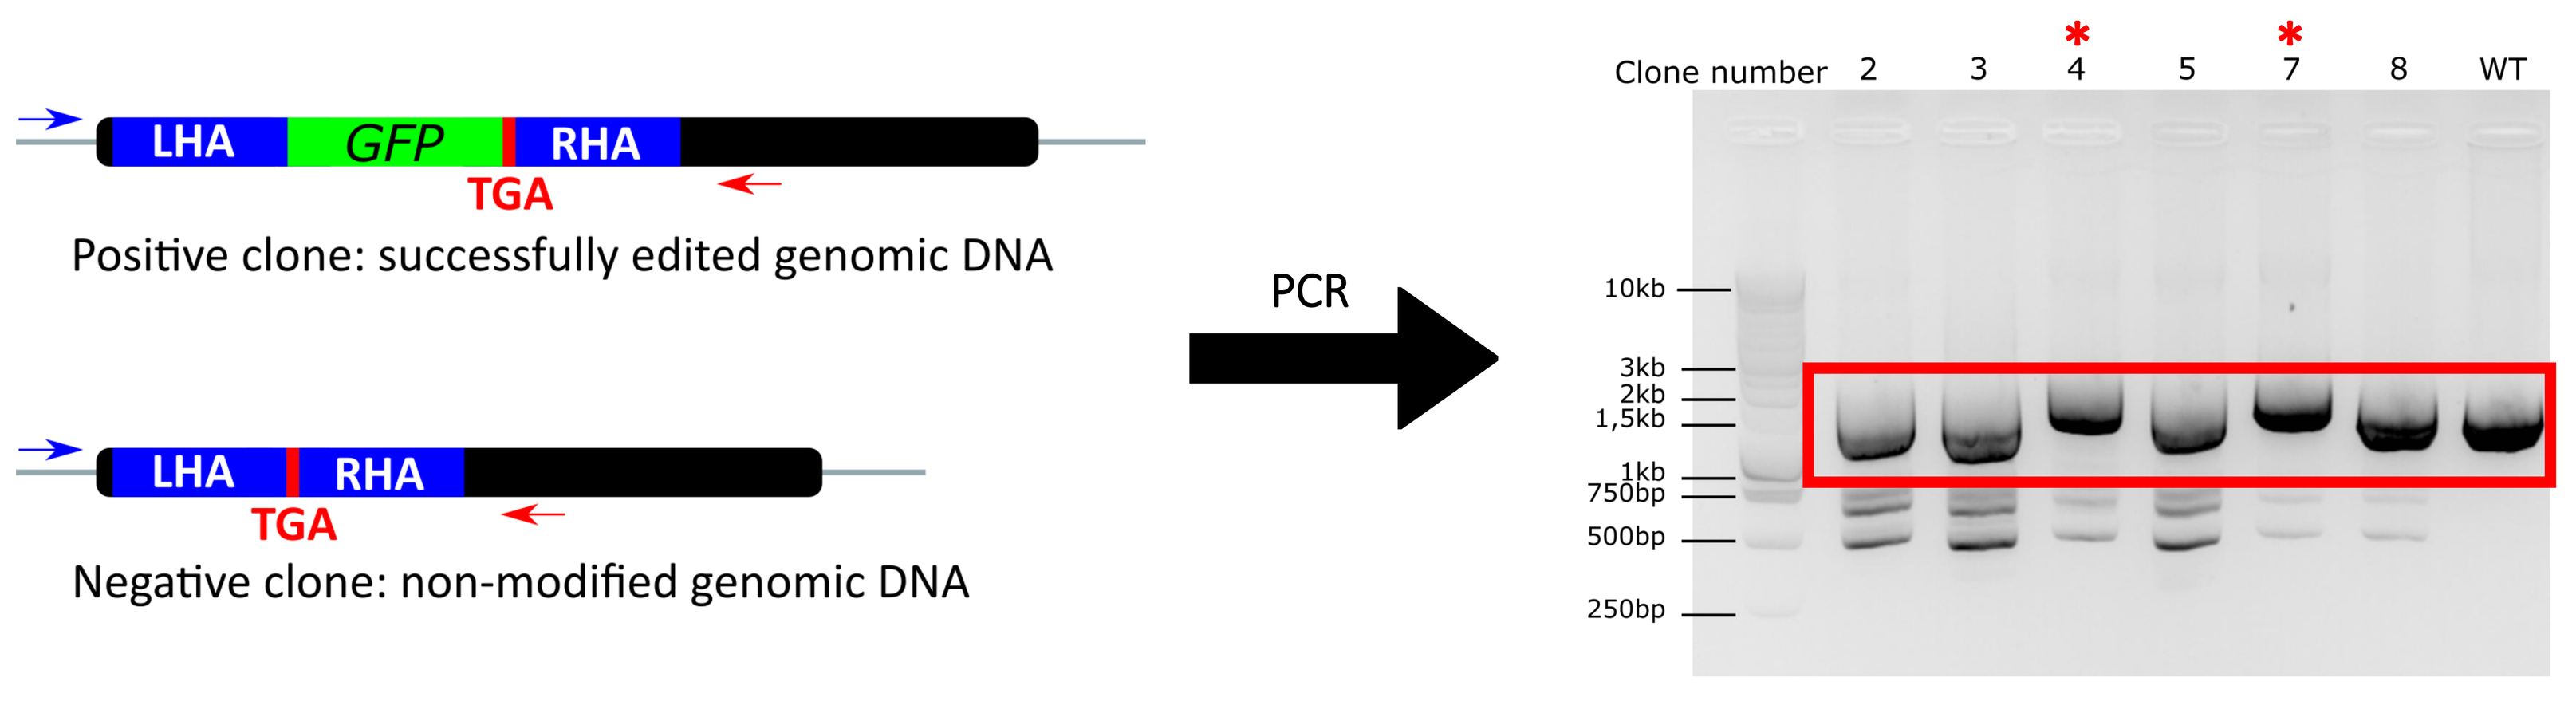
LHA
GFP
RHA
TGA
Positive clone: successfully edited genomic DNA
LHA
RHA
TGA
Negative clone: non-modified genomic DNA
PCR
Clone number 2 3 4 5 7 8 WT
10kb
3kb
2kb
1,5kb
1kb
750bp
500bp
250bp

Figure 4. Control PCR for identifying positive HEK293–HMOX1–GFP clones. The forward and reverse primers are indicated with blue and red arrows, respectively. After PCR, the reaction products were separated on 1.5% agarose gel, and the bands were visualized by staining with GelRed. Two positive clones are indicated by red stars.

2 Inducibility of a positive HEK293–HMOX1–GFP clone upon oxidative stress induction: western blot analysis

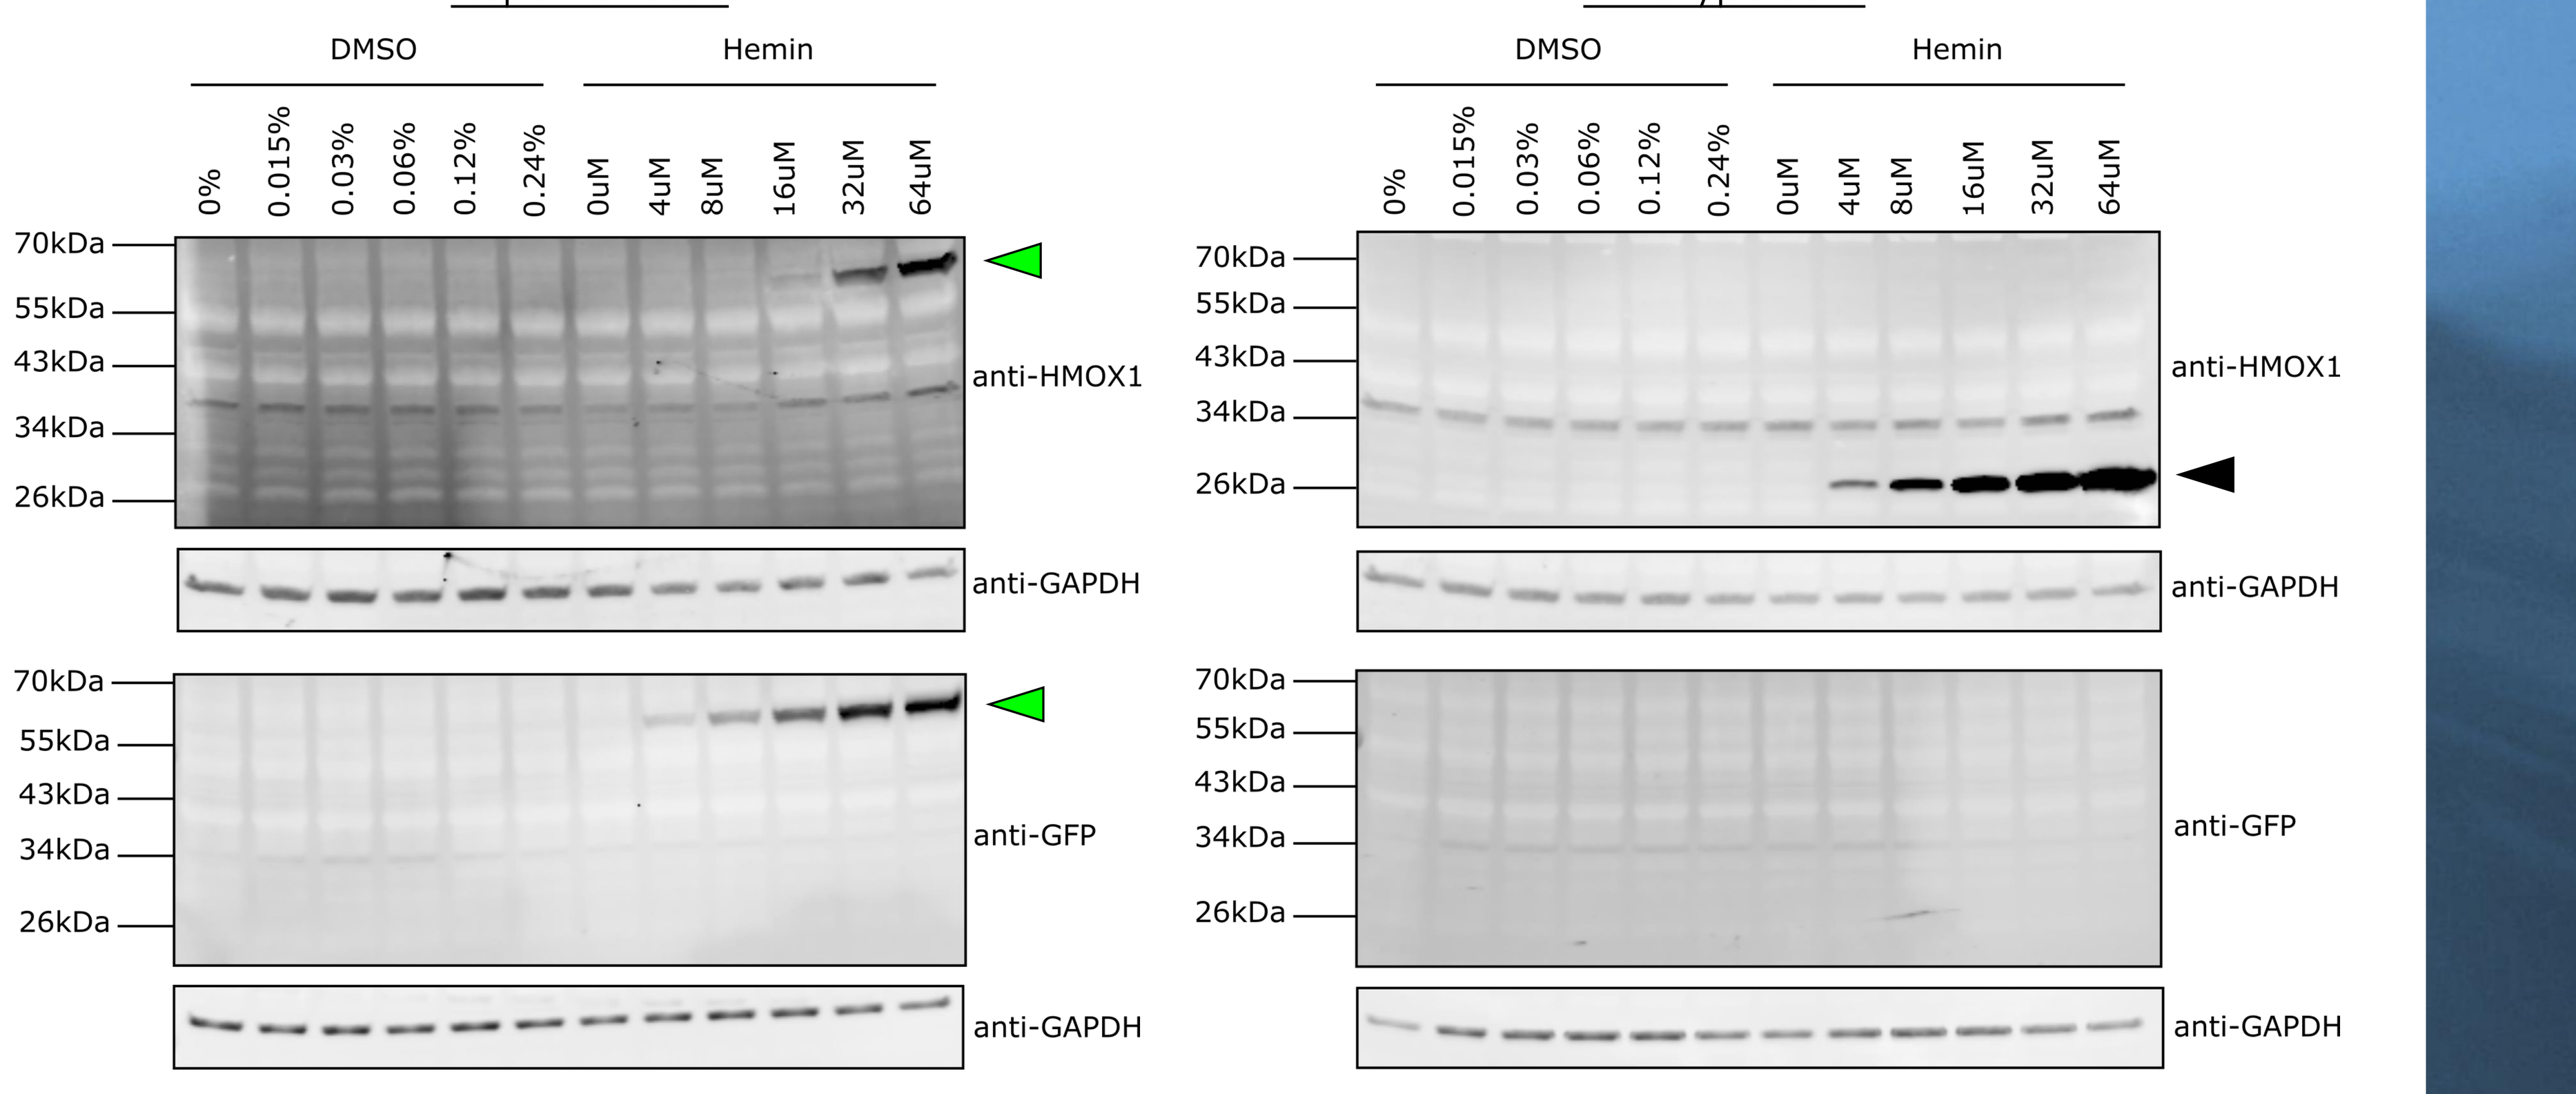
Reporter cells
Wildtype cells
DMSO
Hemin
0%
0.015%
0.03%
0.06%
0.12%
0.24%
0uM
4uM
8uM
16uM
32uM
64uM
70kDa
55kDa
43kDa
34kDa
26kDa
anti-HMOX1
anti-GAPDH
anti-GFP

Figure 5. Inducibility of HMOX1–GFP in the HEK293 reporter cell line upon oxidative stress induction. The fusion protein HMOX1–GFP was detected by western blotting by using anti-GFP and anti-HMOX1 antibodies. HMOX1 and HMOX1–GFP are indicated with black and green triangles, respectively. Anti-GAPDH was used as a loading control.

3 Inducibility of a positive HEK293–HMOX1–GFP clone upon oxidative stress induction: Image analysis

In the case of cigarette smoke extract treatment, direct measurement of the GFP fluorescence signal was unsuccessful because of the intrinsic fluorescence of the cigarette smoke extract in the GFP channel, which resulted in a high background signal. As an alternative, immunocytochemical analysis was performed with a primary anti-GFP antibody and a secondary antibody coupled to Alexa Fluor 647 (Fig. 6).

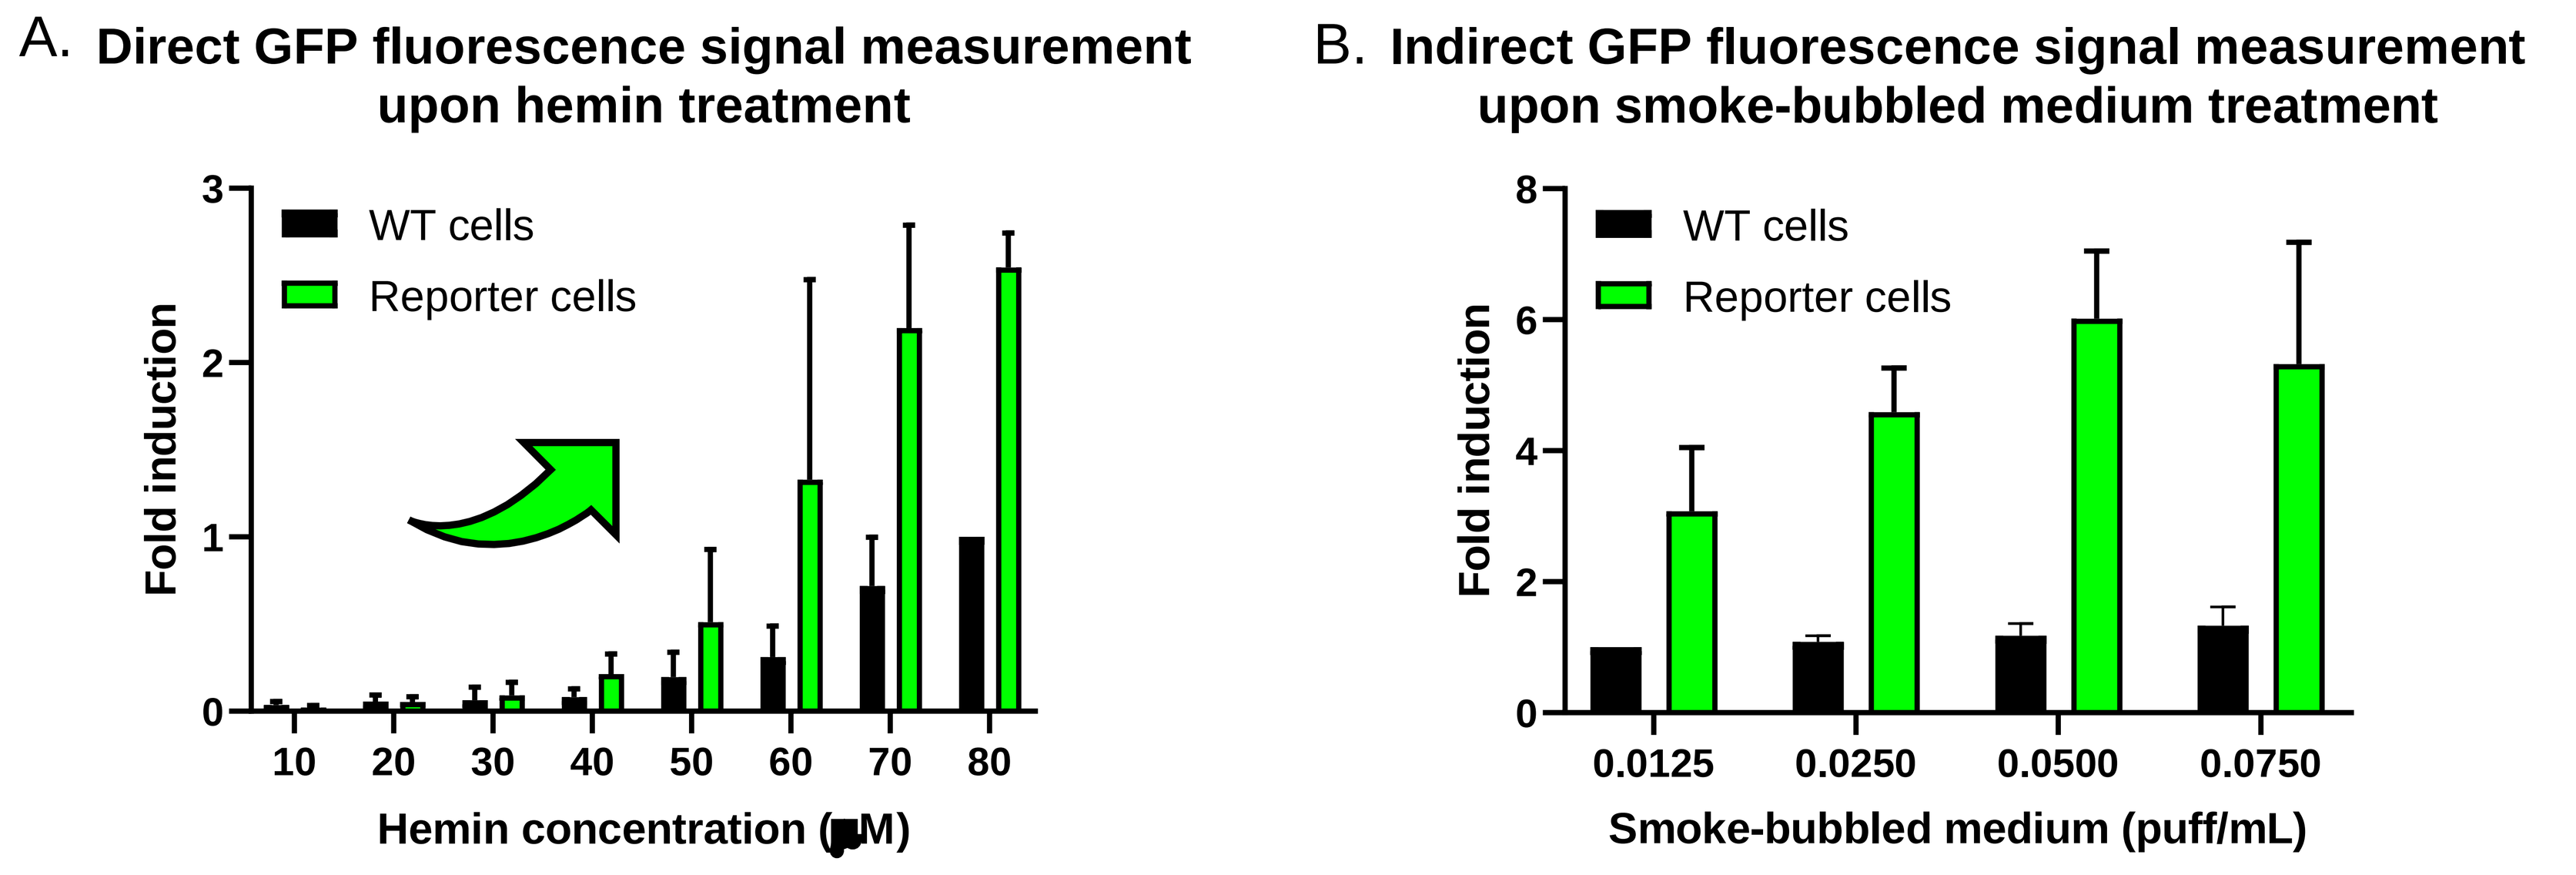
A. Direct GFP fluorescence signal measurement upon hemin treatment
B. Indirect GFP fluorescence signal measurement upon smoke-bubbled medium treatment
WT cells
Reporter cells
Fold induction
10 20 30 40 50 60 70 80
Hemin concentration (µM)
0.0125 0.0250 0.0500 0.0750
Smoke-bubbled medium (puff/mL)

Figure 6. Inducibility of HMOX1–GFP in the reporter cell line upon oxidative stress induction.
A. Direct measurement of GFP fluorescence. B. Indirect measurement of GFP fluorescence (immunocytochemical analysis using an anti-GFP antibody).

Methods

- **CRISPR–Cas9/gRNA plasmid generation:** Guide RNAs (gRNAs) were cloned individually into a CRISPR–Cas9 plasmid by using the technique of fast cloning (Li et al., 2011).
- **DNA repair template generation:** This plasmid was generated by using the Gateway technology. The homology arms were amplified by PCR by using genomic DNA from BEAS-2B cells as a template. The *GFP* gene was amplified from the Gateway™ pcDNA™-DEST47 vector.
- **Electroporation:** HEK293 cells were transfected with 20 µg DNA repair template and 2 µg CRISPR–Cas9/gRNA plasmid by using the Neon™ Transfection System, with the following pulse conditions: 1100 V, 20 ms, and 2 pulses.
- **Clone isolation:** At 24 h post-transfection, 0.5 µg/mL puromycin was added to the cell culture medium. After 1 week of antibiotic selection, limiting dilutions were performed, and clones derived from single cells were isolated and expanded.
- **Cigarette smoke extract generation:** Extracts were generated by bubbling smoke from 1R6F reference cigarettes (University of Kentucky) through minimum essential medium eagle without serum (61–66 puffs per 36 mL). Smoke was generated with a 30-port carousel smoking machine SM-2000 (SM2000; Philip Morris International, Neuchâtel, Switzerland) in accordance with the Health Canada Intense smoking protocol (Health Canada, 1999).
- **Evaluation of reporter cells' inducibility:** For western blot analysis, cells were seeded in a 24-well plate and treated with hemin or DMSO for 17 hours. The cells were then collected, and proteins were extracted by using the M-PER™ Mammalian Protein Extraction Reagent. Protein samples were analyzed by western blotting by using anti-GFP, anti-HMOX1, and anti-GAPDH antibodies. For imaging analysis, cells were seeded in a 96-well plate and treated with hemin or cigarette smoke extract for 17 to 24 hours. The cells were then fixed with 4% formaldehyde and stained with 1 µg/mL Hoechst 33342 in PBS. The cells were imaged, either directly or after staining with an anti-GFP antibody, by using the Cellinsight ™ CX7 platform.

References

- Health Canada (1999). Determination of Tar, Water, Nicotine and Carbon Monoxide in Mainstream Tobacco Smoke.
- Labun, K., Montague, T. G., Krause, M., Torres Cleuren, Y. N., Tjeldnes, H., & Valen, E. (2019). CHOPCHOP v3: expanding the CRISPR web toolbox beyond genome editing. *Nucleic Acids Res*, *47*(W1), W171-W174. https://doi.org/10.1093/nar/gkz365
- Li, C., Wen, A., Shen, B., Lu, J., Huang, Y., & Chang, Y. (2011). FastCloning: a highly simplified, purification-free, sequence- and ligation-independent PCR cloning method. *BMC Biotechnol*, *11*, 92. https://doi.org/10.1186/1472-6750-11-92

11TH WORLD CONGRESS ON ALTERNATIVES AND ANIMAL USE IN THE LIFE SCIENCES (Virtual Meeting), 23 August – 2 September 2021

Competing Financial Interest: The research described in this poster was sponsored by Philip Morris International .