Systems Toxicology Assessment of a Candidate Modified Risk Tobacco Product Compared to a Combustible Cigarette

Ignacio Gonzalez-Suarez[1], Florian Martin[1], Diego Marescotti[1], Emmanuel Guedj[1], Stefano Acali[1], Stephanie Johne[1], Remi Dulize[1], Karine Baumer[1], Dariusz Peric[1], Didier Goedertier[1], Stefan Frentzel[1], Nikolai V. Ivanov[1], Carole Mathis[1], A. Wallace Hayes[2], Julia Hoeng[1] & Manuel C. Peitsch[1]

1. Philip Morris International R&D, Philip Morris Products S.A., Quai Jeanrenaud 5, 2000 Neuchâtel, Switzerland (part of Philip Morris International group of companies)

2. Spherix Consulting, Rockville, MD, USA and a Member of the Scientific Advisory Board of PMI R&D

P135

ABSTRACT

Cigarette smoke (CS) increases the risk for respiratory and other diseases[1] and cessation is the most effective approach to minimize the risk of smoking-related disease. However, for those unwilling to quit smoking, modified risk tobacco products (MRTP) may be useful to lower disease burden.[2] Cigarettes burn at temperatures around 900ºC when a puff is taken,[3] resulting in partial combustion of the tobacco leaf and the generation of smoke. CS is a complex mixture with more than 8,000 identified chemicals,[4] many of which are considered toxic or carcinogenic[5] and suspected to be responsible for tobacco-related diseases. The tobacco heating system (THS2.2) is a candidate MRTP composed of an electronic holder where the tobacco stick is inserted and heated by an electronically-controlled heating blade. Heating the tobacco generates an aerosol mainly composed of water and glycerol that contains lower levels of harmful and potentially harmful constituents (HPHCs).

We initially performed a chemical characterization of mainstream THS 2.2 aerosol. Moreover, we investigated the biological impact of THS 2.2, compared to the 3R4F reference cigarette in normal primary human bronchial epithelial cells. Cells were exposed to 3 different smoke/aerosol fractions: an aqueous extract generated by bubbling mainstream 3R4F smoke or THS2.2 aerosol through PBS, total particulate matter (TPM) and gas-vapor phase (the substance that passes through the filter during TPM collection). Multiple toxicity endpoints were measured *via* real-time cellular analysis and high-content screening. The study was complemented by gene expression analysis, followed by a computational approach to identify and quantify perturbed molecular pathways.

MATERIAL AND METHODS

Real-time cell analyzer

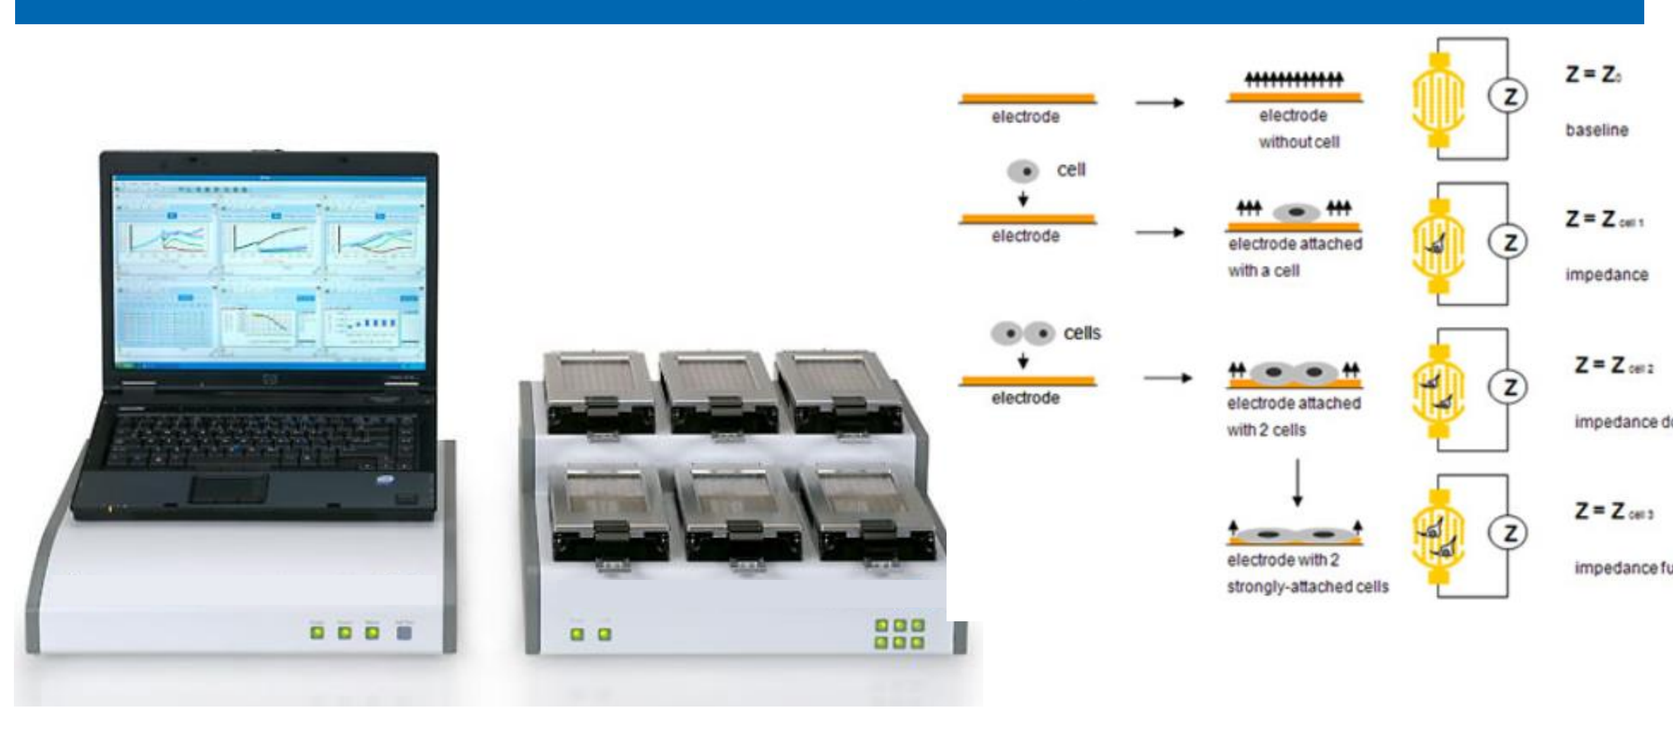

electrode
electrode without cell
Z = Z0
baseline
cell
electrode attached with a cell
Z = Z cell 1
impedance
cells
electrode attached with 2 cells
Z = Z cell 2
impedance doubly
electrode with 2 strongly-attached cells
Z = Z cell 3
impedance further

High Content Screening

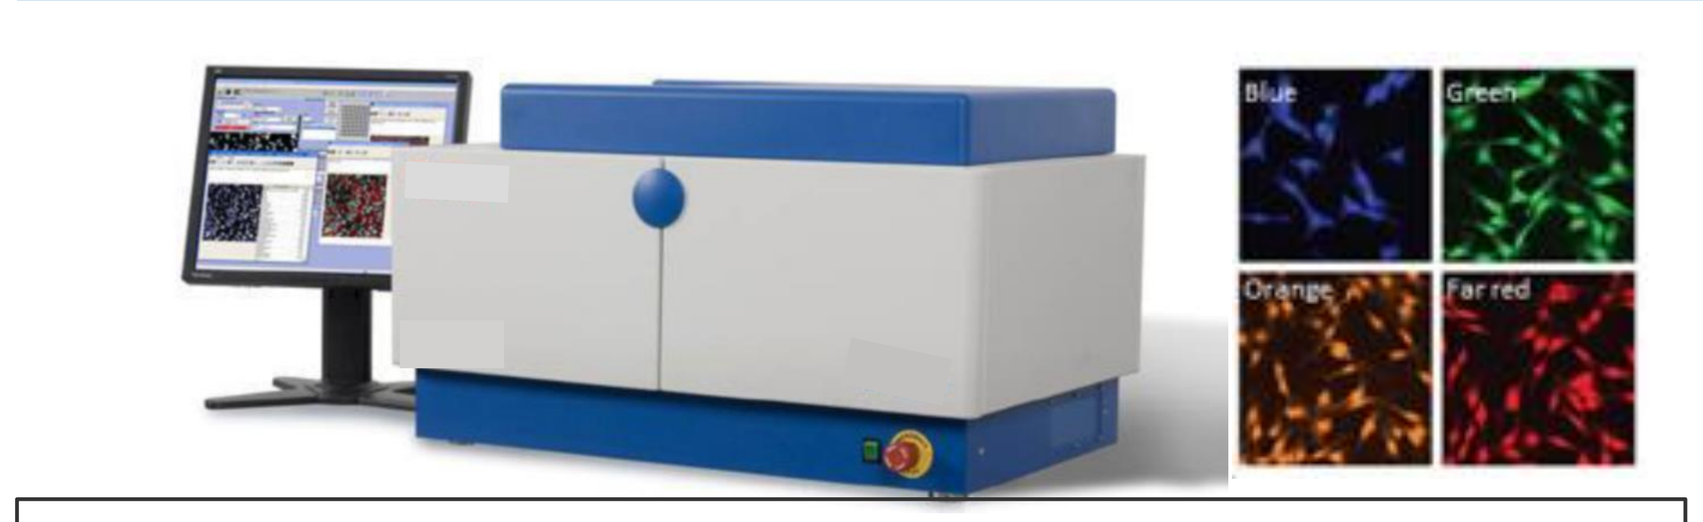

Blue
Green
Orange
Far red

- Cell count
- DNA damage
- Stress kinase
- Oxidative Stress
- Glutathione Content
- Cell cycle
- Caspase 3/7
- Cytochrome C release
- Mitochondrial potential
- Mitochondrial mass
- Cell membrane permeability

Smoke / Aerosol Generation

- Chemical characterization was performed on mainstream 3R4F smoke and THS 2.2 aerosol.
- NHBE cells were exposed to 3R4F smoke THS 2.2 aerosol fractions (see figure below)

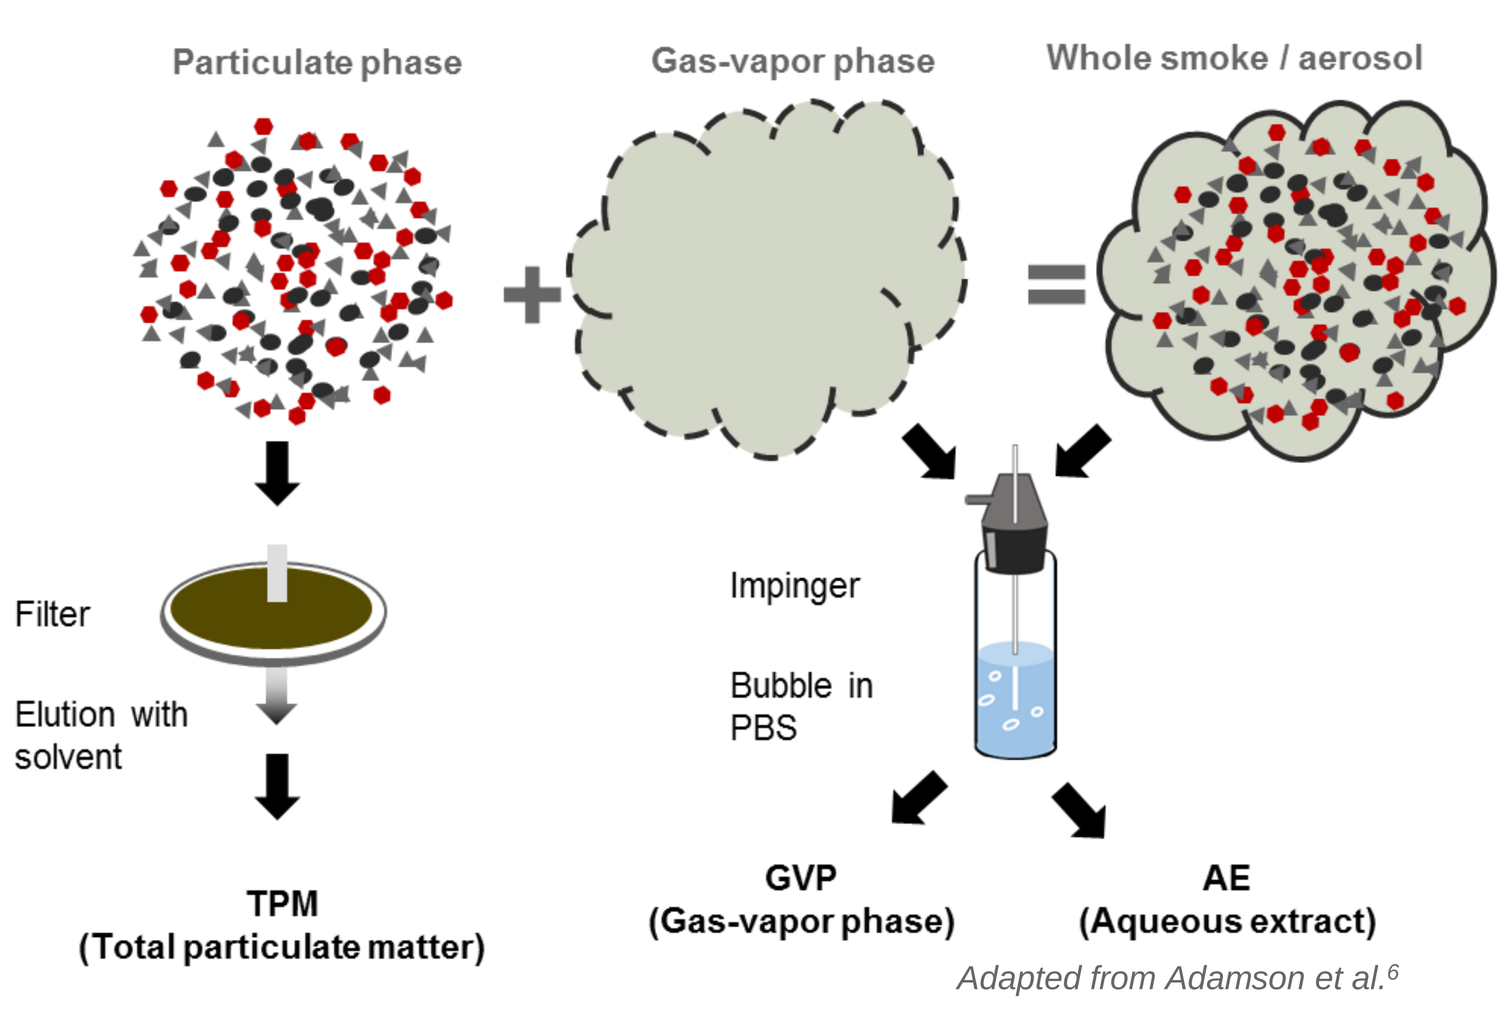

Particulate phase
Gas-vapor phase
Whole smoke / aerosol
Filter
Elution with solvent
Impinger
Bubble in PBS
TPM
(Total particulate matter)
GVP
(Gas-vapor phase)
AE
(Aqueous extract)

Adapted from Adamson et al.[6]

Gene Expression Analysis

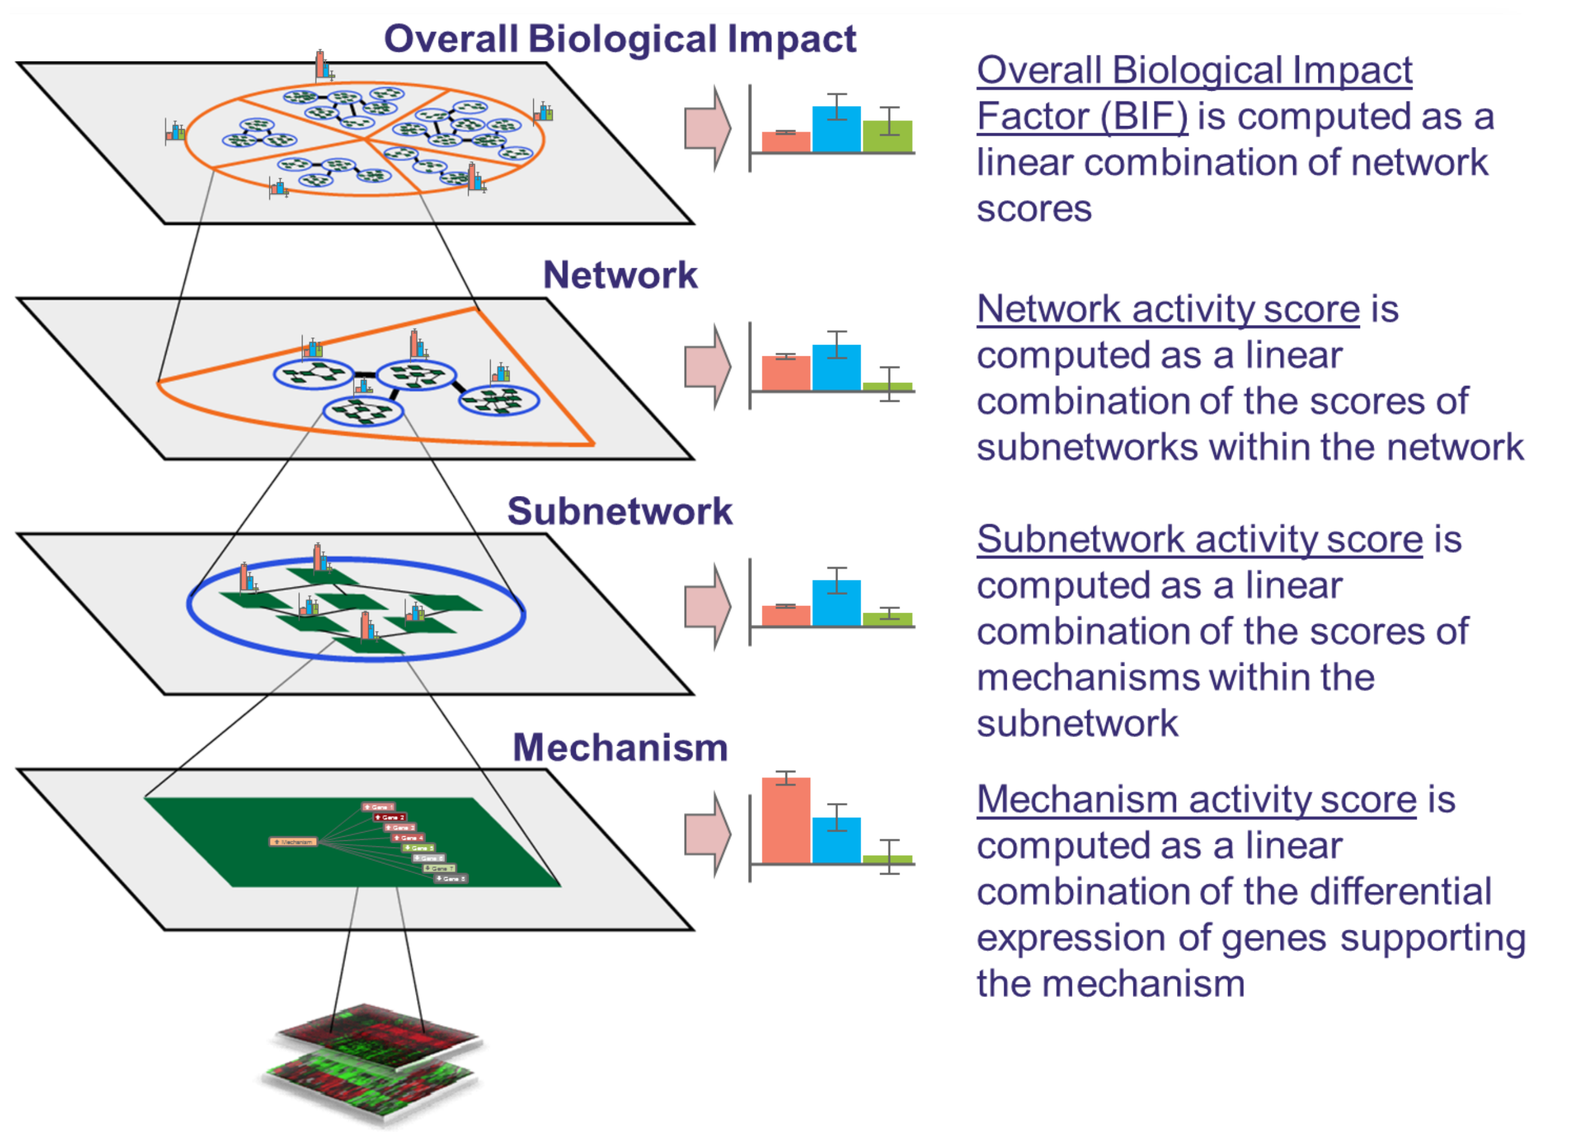

Overall Biological Impact
Network
Subnetwork
Mechanism
Overall Biological Impact Factor (BIF) is computed as a linear combination of network scores
Network activity score is computed as a linear combination of the scores of subnetworks within the network
Subnetwork activity score is computed as a linear combination of the scores of mechanisms within the subnetwork
Mechanism activity score is computed as a linear combination of the differential expression of genes supporting the mechanism

CHEMICAL CHARACTERIZATION

Mainstream THS 2.2 aerosol has lower HPHC levels compared to mainstream 3R4F smoke.

	Parameters	Units	3R4F	THS 2.2	THS 2.2 Vs. 3R4F (%)
1	NFDPM	mg/cig	31.2 ± 0.6	10.3 ± 0.3	-67%
2	Glycerol	mg/cig	2.42 ± 0.04	4.63 ± 0.26	+91%
3	TPM	mg/cig	49 ± 1.5	48.2 ± 0.8	-2%
4	Nicotine	mg/cig	1.89 ± 0.05	1.32 ± 0.05	-30%
5	Water	mg/cig	15.8 ± 0.9	36.5 ± 1	+131%
6	Carbon monoxide	mg/cig	32.8 ± 0.7	0.531 ± 0.021	-98%
7	1,3-butadiene	µg/cig	63.8 ± 1.1	0.294 ± 0.013	-99.9%
8	Isoprene	µg/cig	798 ± 15	2.35 ± 0.12	-99.9%
9	Formaldehyde	µg/cig	56.5 ± 3.8	5.53 ± 0.22	-90%
10	Acetaldehyde	µg/cig	1555 ± 38	219 ± 10	-86%
11	Acetone	µg/cig	736 ± 41	40.7 ± 1.9	-94%
12	Acrolein	µg/cig	154 ± 6	11.3 ± 0.7	-93%
13	Propionaldehyde	µg/cig	125 ± 5	14.5 ± 0.7	-88%
14	Crotonaldehyde	µg/cig	68.8 ± 4.5	4.14 ± 0.07	-94%
15	Methyl-ethyl-ketone	µg/cig	187 ± 9	7.18 ± 0.37	-96%
16	Butyraldehyde	µg/cig	88.4 ± 3.4	26.1 ± 0.7	-70%
17	Acetamide	µg/cig	13.9 ± 0.2	4.02 ± 0.06	-71%
18	Acrylamide	µg/cig	4.83 ± 0.08	1.73 ± 0.04	-64%
19	Acrylonitrile	µg/cig	31.9 ± 0.6	0.258 ± 0.013	-99%
20	Ethylene oxide	µg/cig	29.4 ± 0.6	0.201 ± 0.004	-99%
21	Propylene oxide	µg/cig	1.32 ± 0.04	0.148 ± 0.006	-89%
22	Nitrobenzene	ng/cig	8.62 ± 0.35	<0.188* ± *	-98%
23	1-Aminonaphthalene	ng/cig	20.8 ± 0.4	0.077* ± *	-99%
24	2-Aminonaphthalene	ng/cig	11 ± 0.2	0.046 ± 0.002	-99%
25	3-Aminobiphenyl	ng/cig	3.77 ± 0.15	<0.032* ± *	-99%
26	4-Aminobiphenyl	ng/cig	3.26 ± 0.04	<0.051* ± *	-99%
27	o-Toluidine	ng/cig	85.5 ± 0.8	1.26 ± 0.06	-99%
28	Benzidine	ng/cig	<0.017* ± *	<0.014* ± *	N/A
29	Pyridine	µg/cig	36.1 ± 0.7	7.54 ± 0.08	-79%
30	Quinoline	µg/cig	0.513 ± 0.007	<0.012* ± *	-98%
31	Vinyl chloride	ng/cig	96.7 ± 0.6	<3.54* ± *	-96%
32	Ammonia	µg/cig	39.3 ± 1	14.2 ± 0.3	-64%
33	Nitrogen oxide	µg/cig	491 ± 12	16.8 ± 0.7	-97%
34	Nitrogen oxides	µg/cig	537 ± 14	17.3 ± 0.8	-97%
35	Hydrogen cyanide	µg/cig	493 ± 24	4.81 ± 0.11	-99%
36	Benzene	µg/cig	97.6 ± 1.5	0.649 ± 0.023	-99%
37	Styrene	µg/cig	24.5 ± 0.4	0.608 ± 0.018	-98%
38	Toluene	µg/cig	188 ± 4	2.59 ± 0.14	-99%
39	NNN	ng/cig	309 ± 13	17.2 ± 0.4	-94%
40	NAT	ng/cig	318 ± 23	20.5 ± 0.1	-94%
41	NAB	ng/cig	33.7 ± 2.7	<3.15* ± *	-90%
42	NNK	ng/cig	266 ± 5	6.67 ± 0.19	-97%
43	Phenol	µg/cig	13.6 ± 0.3	1.16 ± 0.04	-91%
44	o-Cresol	µg/cig	4.47 ± 0.05	0.069 ± 0.003	-98%
45	m-Cresol	µg/cig	3.03 ± 0.02	0.029 ± 0.001	-99%
46	p-Cresol	µg/cig	9.17 ± 0.14	0.072 ± 0.003	-99%
47	Catechol	µg/cig	91.4 ± 1.8	16.3 ± 0.5	-82%
48	Resorcinol	µg/cig	1.85 ± 0.02	0.041 ± 0.001	-98%
49	Hydroquinone	µg/cig	83.1 ± 1.7	8.1 ± 0.15	-90%
50	Benzo[a]pyrene	ng/cig	14.2 ± 0.1	<1.00* ± *	-93%
51	Pyrene	ng/cig	87.3 ± 0.8	<5.00* ± *	-94%
52	Benzo(a)anthracene	ng/cig	28 ± 0.2	1.45 ± 0.04	-95%
53	Dibenzo(a,h)anthracene	ng/cig	1.7 ± 0.03	<0.100* ± *	-94%
54	Arsenic	ng/cig	8.51 ± 0.11	<1.13* ± *	-87%
55	Cadmium	ng/cig	161 ± 1	<0.350* ± *	-99%
56	Chromium	ng/cig	<0.550* ± *	<0.550* ± *	N/A
57	Lead	ng/cig	37 ± 0.2	<3.35* ± *	-91%
58	Mercury	ng/cig	4.8 ± 0.04	1.17 ± 0.02	-76%
59	Nickel	ng/cig	<0.550* ± *	<0.550* ± *	N/A
60	Selenium	ng/cig	1.62 ± 0.1	<0.550* ± *	-66%

Table 1. Characterization of 3R4F smoke and THS 2.2 aerosol. Values represent AVG ± StDEV deviation of 4 independent determinations. Top right column represents % change in THS 2.2 compared to 3R4F. * Values below limit of quantification (LOQ). In those cases, LOQ was used to calculate % of change. If both items were below LOQ, % change could not be calculated (N/A). NNN (N-Nitrosonornicotine), NAT (N-Nitrosoanatabine), NAB (N-Nitrosoanabasine), NNK (4-(N-Nitrosomethylamino)-1-(3-pyridyl)-1-butanone).[7]

RESULTS

THS 2.2 aerosol fractions caused less NHBE cell cytotoxicity compared to 3R4F smoke fractions.

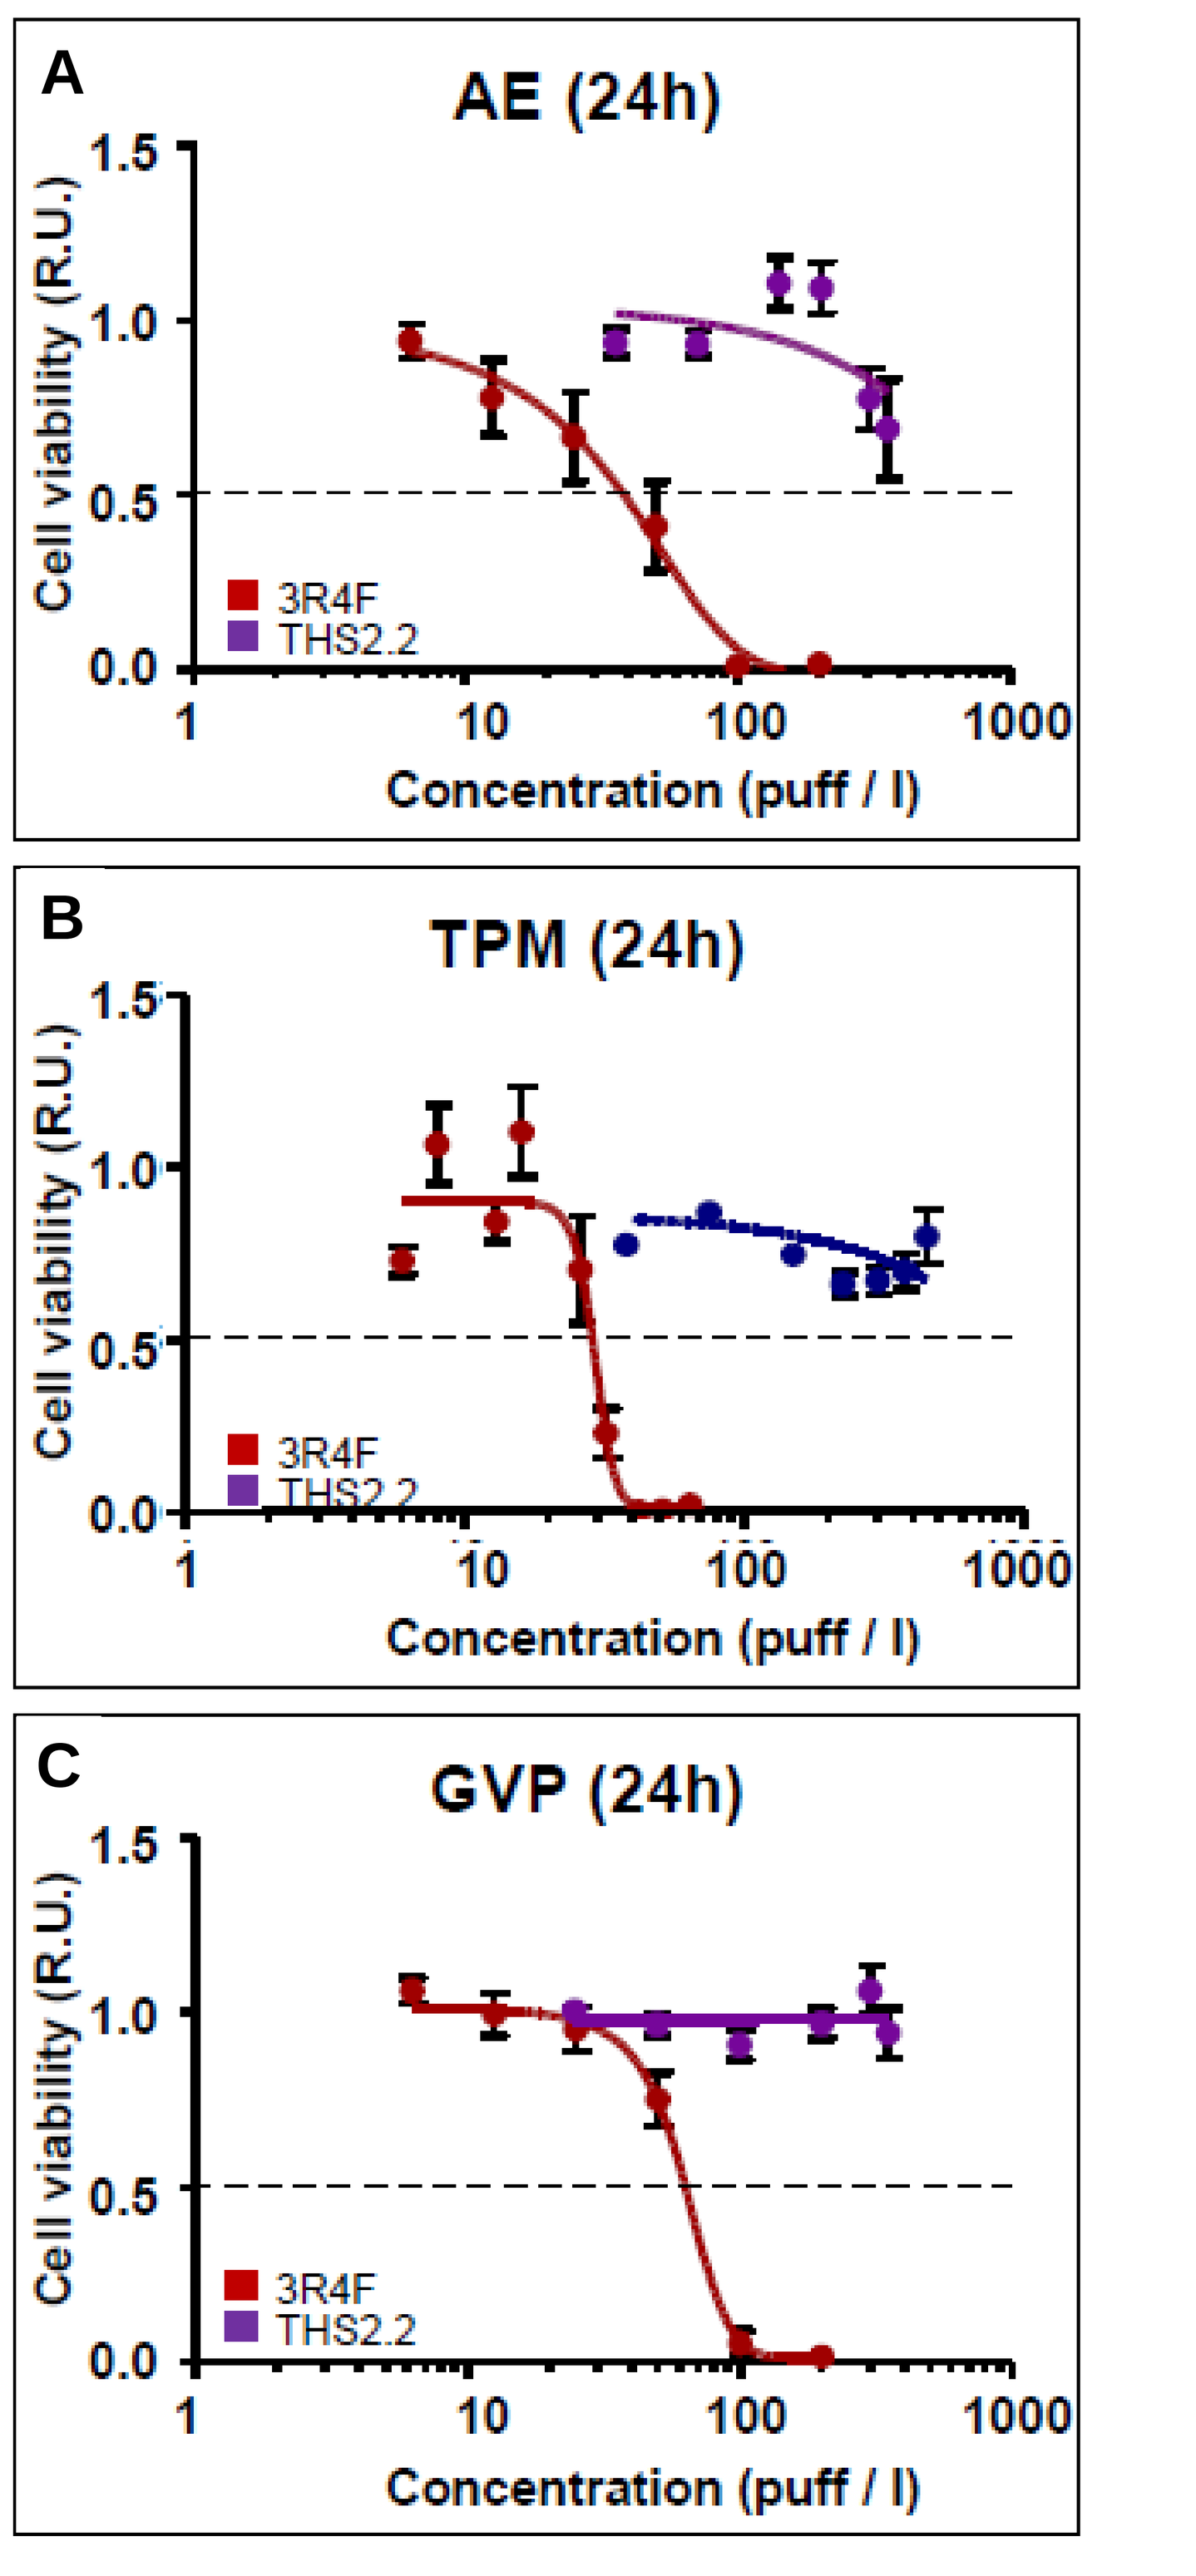

A
AE (24h)
B
TPM (24h)
C
GVP (24h)
Cell viability (R.U.)
3R4F
THS2.2
Concentration (puff / l)

Figure 1. **Cell viability.** NHBE cells were exposed 24h to AE (A), TPM (B) or GVP (C) from 3R4F smoke or THS 2.2 aerosol. Values represent average ± SEM of at least three independent experiments. Dotted line indicates 50% cell viability.

THS 2.2 aerosol fractions exhibit reduced toxicity in NHBE cells compared to 3R4F smoke fractions.

Endpoint	Exposure	s/aPBS		TPM		GVP	
		3R4F	THS 2.2	3R4F	THS 2.2	3R4F	THS 2.2
Cell Loss	4h	-	-	-	-	-	-
	24h	100	200[a]	33	-	200	-
p-H2AX	4h	*200[b]*	-	52	-	*200[b]*	-
	24h	*200[b]*	-	8	226	*200[b]*	-
p-cJun	4h	-	-	*33[b]*	-	-	-
	24h	100	200[a]	33	-	100	-
ROS formation	4h	-	-	N/A	-	-	-
	24h	100[a]	-	N/A	350	*200[b]*	-
GSH content	4h	50	-	33[a]	-	50	350
	24h	100	200[a]	42	-	200	-
Cell Cycle	24h	13	140	8	150	25	200
Caspase 3/7	4h	-	-	N/A	380[a]	-	-
	24h	-	-	N/A	280	200[a]	300[a]
Cytochrome C release	4h	-	-	-	-	-	-
	24h	100	280[a]	42	380*	*200[b]*	-
Cell membrane permeability	4h	100	*350[b]*	65	-	200[a]	-
	24h	100	-	8	150	100	350[a]
Mitochondrial membrane potential	4h	-	-	16	380[a]	-	-
	24h	100	280	62	280	200	-
Mitochondrial mass	4h	50[a]	-	-	-	200[a]	-
	24h	*200[b]*	-	-	280	200[a]	-

Table 2. Summary of HCS results. Only endpoints for which a positive response was observed in at least one experimental condition are listed. Values indicate the minimum concentration (puffs/L) at which at least a 2-fold increase in signal above vehicle was observed (50% decrease for cell count, GSH content and mitochondrial membrane potential). Cell cycle values represent the minimum concentration at which a 50% decrease in the percentage of cells in S-phase was observed. [a] indicates a weak response (1.5-2.0 fold increase in signal above vehicle or 30-50% decrease in cell count, GSH and mitochondrial membrane potential). [b] indicates that the response was not dose-dependent. Caspase 3/7 activity and ROS formation could not be measured in 3R4F TPM because of interferences in fluorescence emission (N/A).

Exposure to THS 2.2 aerosol has a lower biological impact in NHBE cell transcriptome compared to 3R4F smoke.

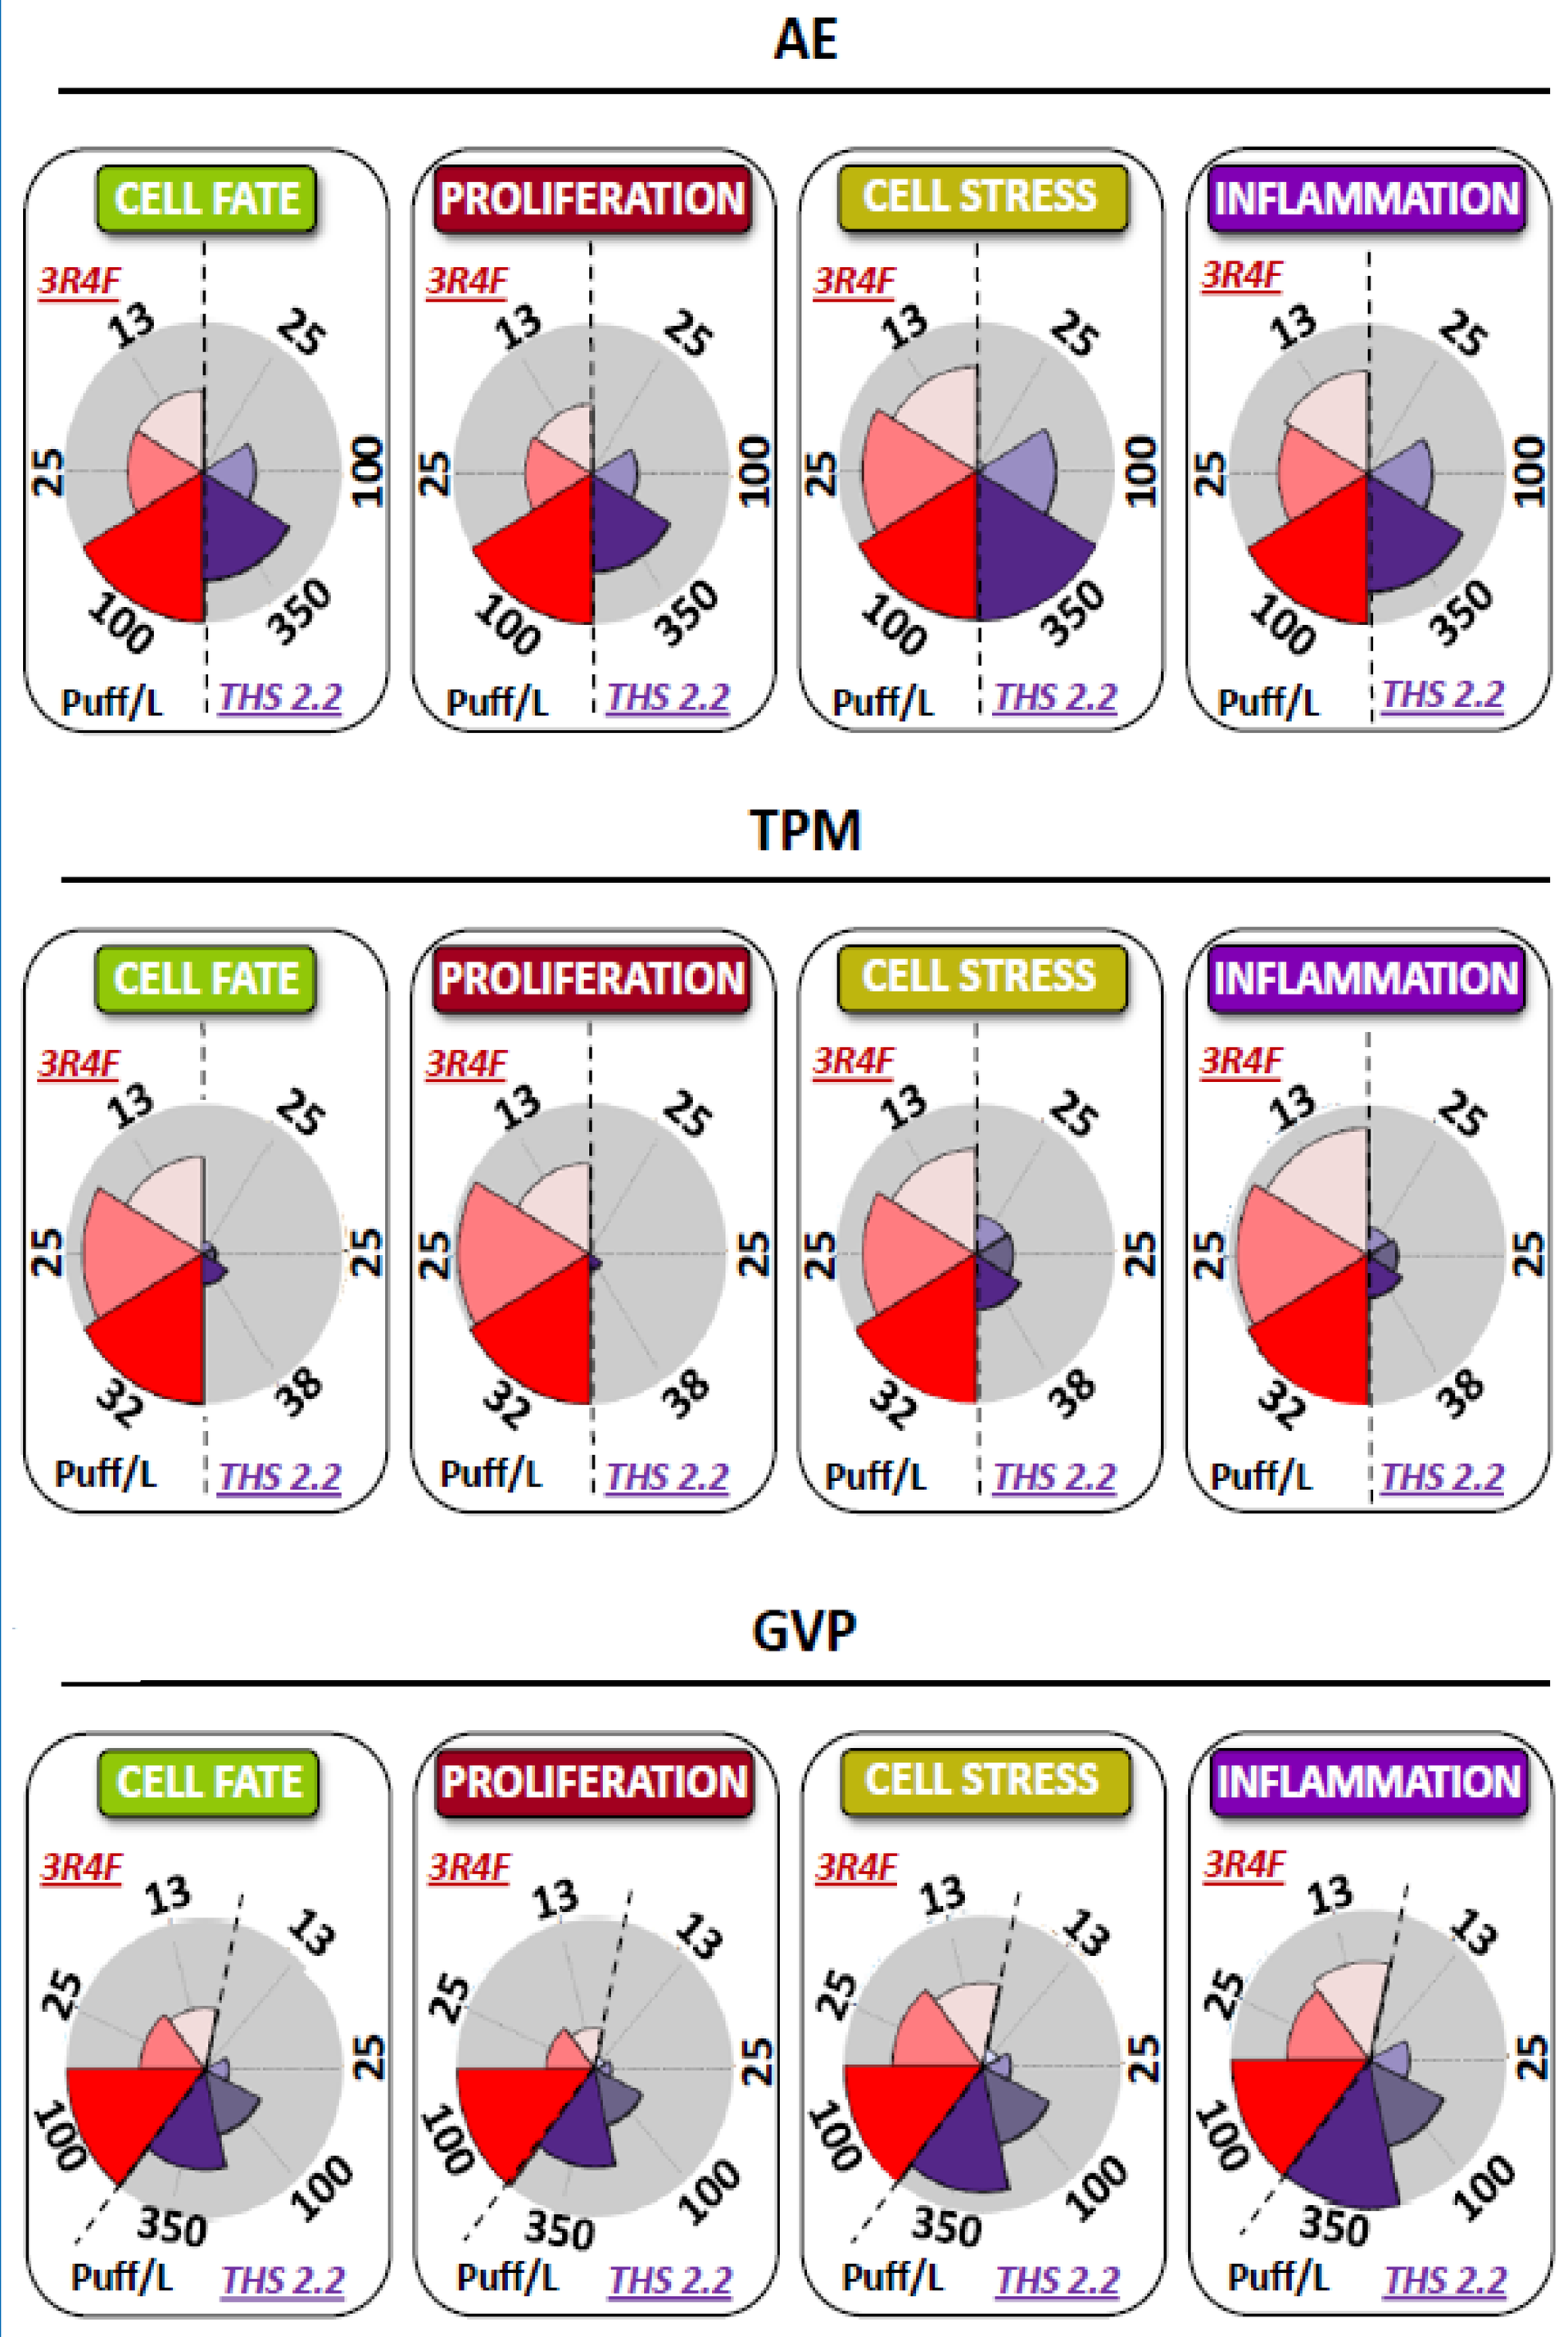

AE
TPM
GVP
CELL FATE
PROLIFERATION
CELL STRESS
INFLAMMATION
3R4F
THS 2.2
Puff/L

Figure 3. Biological impacts on the network models after 4h exposure to AE, TPM or GVP fractions from 3R4F smoke or THS 2.2 aerosol. The pie chart represents the distribution of the sum of contributions for each network across all treatment groups. The surface area of the different segments within each plot is normalized to the dose showing the maximum level of network perturbation, which is used as a reference. The sum of all contributions for each treatment is 100%.

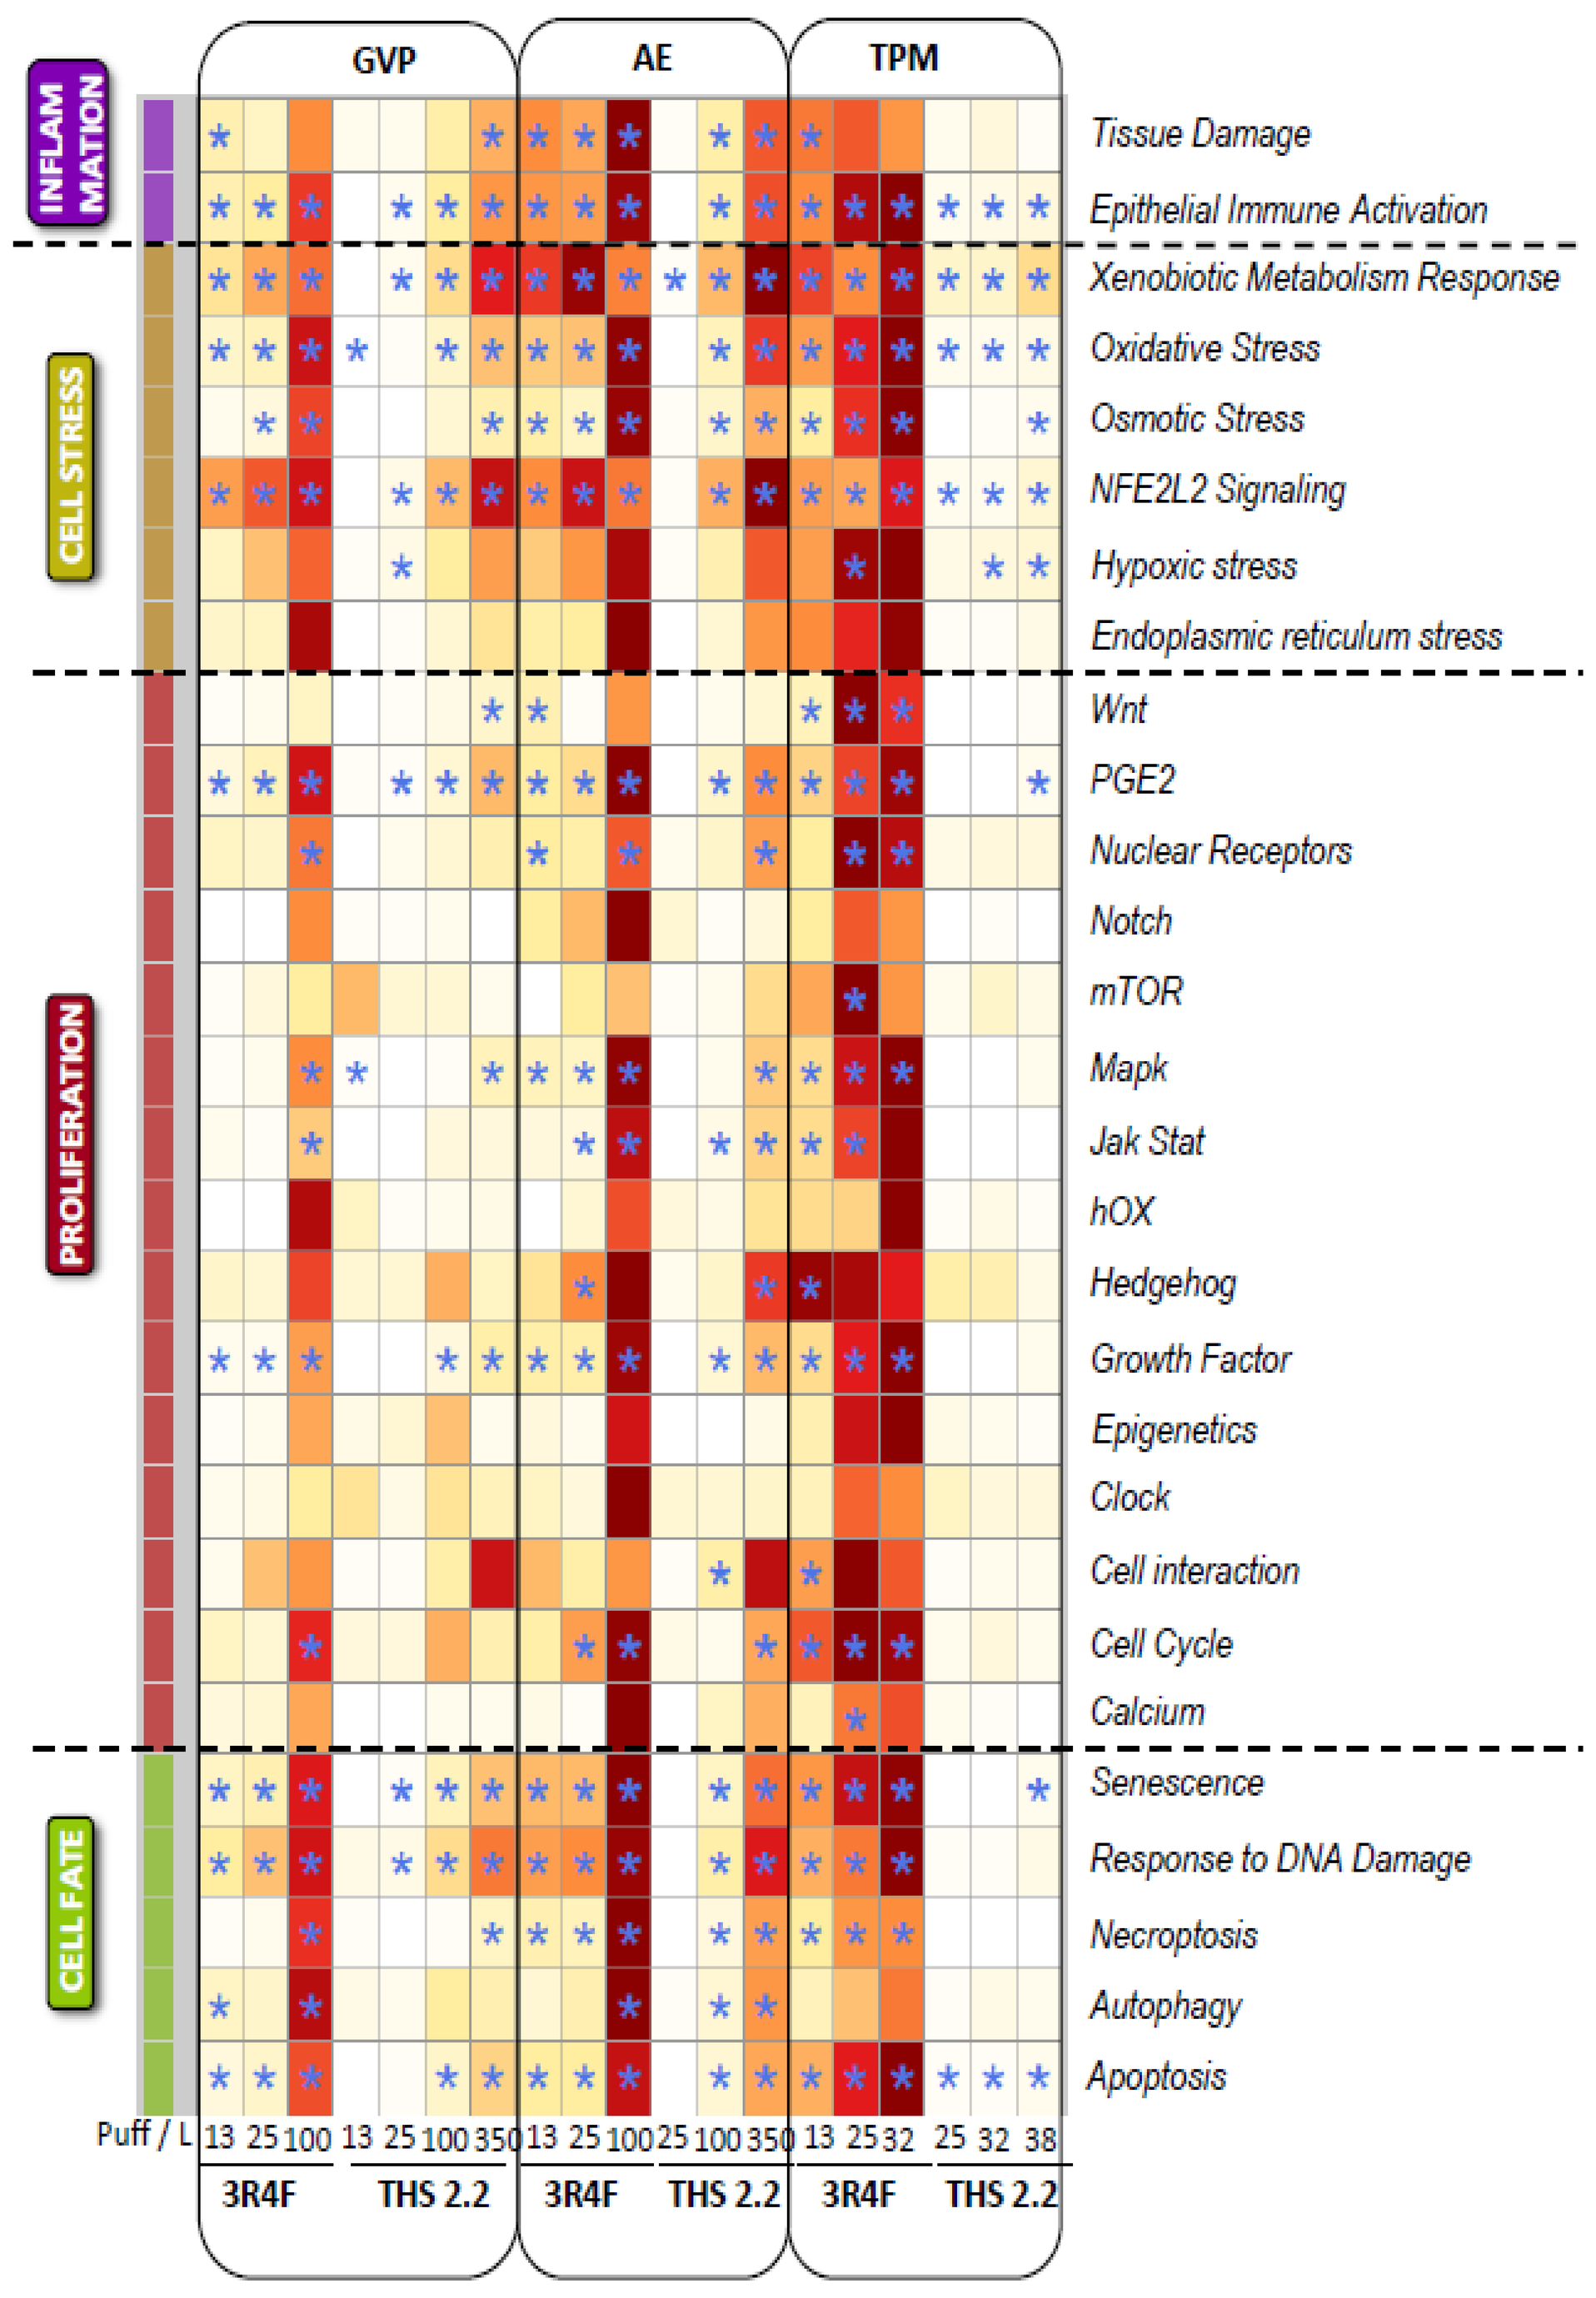

GVP
AE
TPM
INFLAMMATION
CELL STRESS
PROLIFERATION
CELL FATE
Tissue Damage
Epithelial Immune Activation
Xenobiotic Metabolism Response
Oxidative Stress
Osmotic Stress
NFE2L2 Signaling
Hypoxic stress
Endoplasmic reticulum stress
Wnt
PGE2
Nuclear Receptors
Notch
mTOR
Mapk
Jak Stat
hOX
Hedgehog
Growth Factor
Epigenetics
Clock
Cell interaction
Cell Cycle
Calcium
Senescence
Response to DNA Damage
Necroptosis
Autophagy
Apoptosis
Puff / L
3R4F
THS 2.2

Figure 4. NPA heatmaps for NHBE cells exposed to 3R4F smoke and THS 2.2 aerosol fractions. Darker colors indicate higher NPA scores. Significantly perturbed networks are indicated as *.

Exposure to THS 2.2 aerosol has a lower effect on NHBE cell transcriptome compared to 3R4F smoke

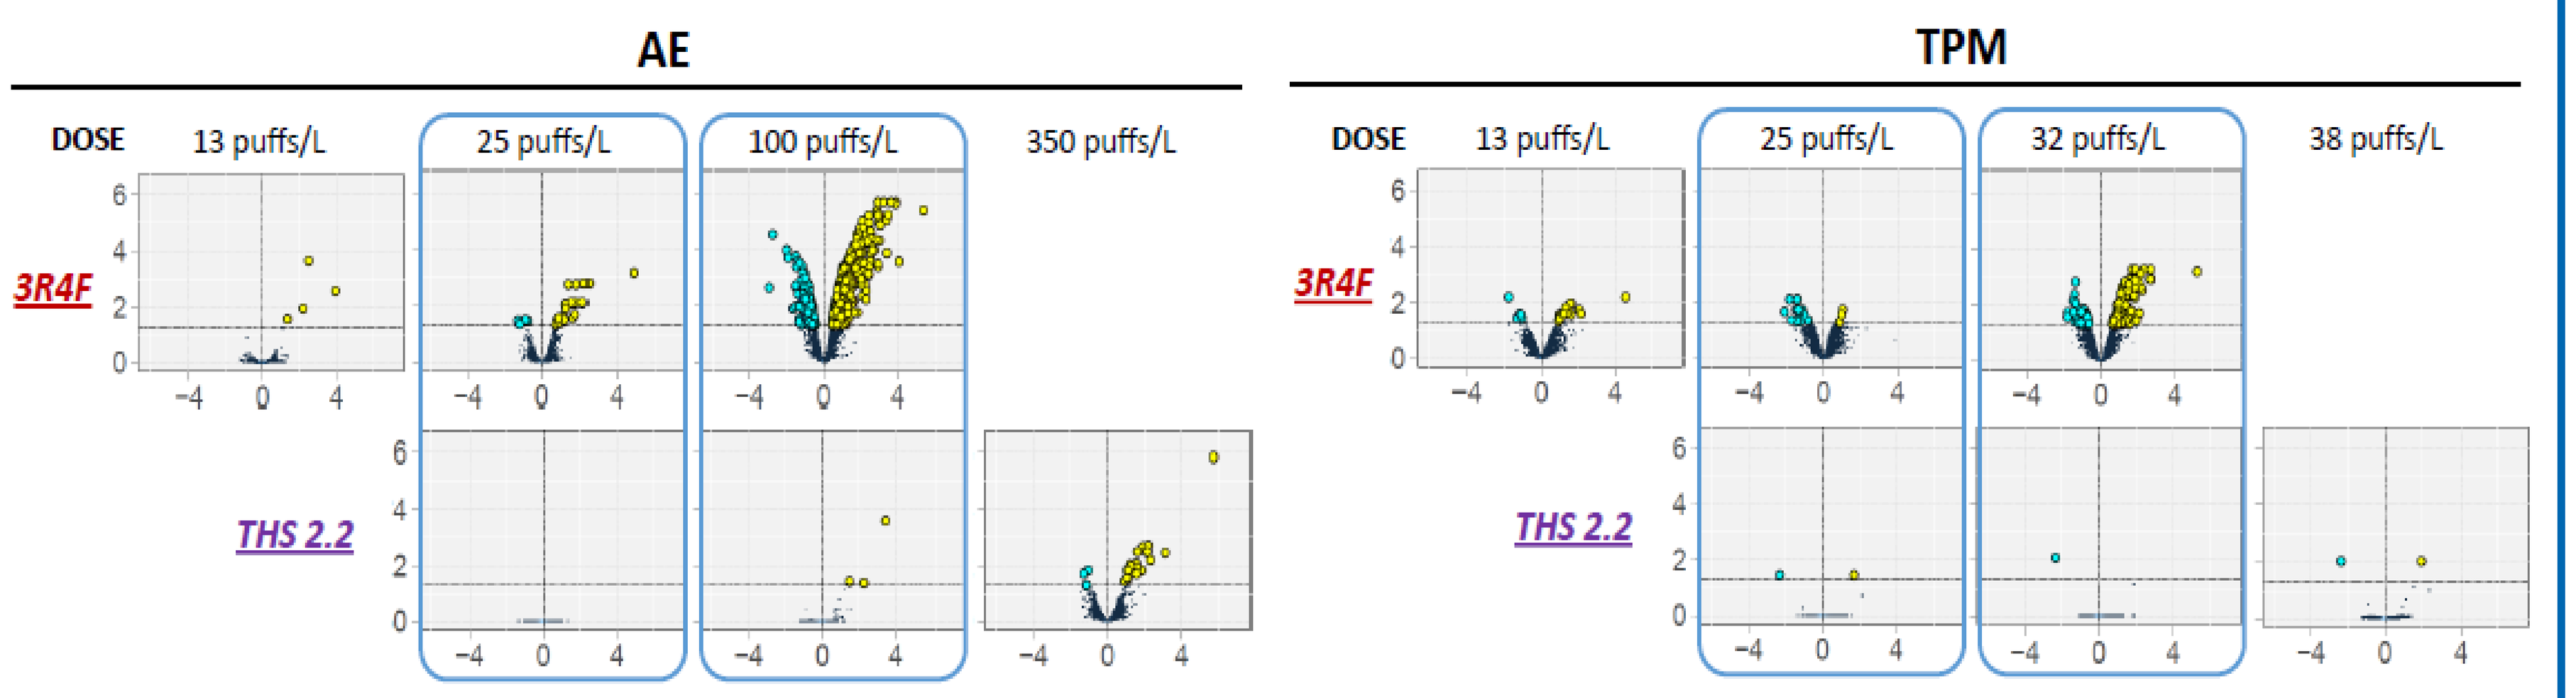

AE
TPM
DOSE
13 puffs/L
25 puffs/L
100 puffs/L
350 puffs/L
32 puffs/L
38 puffs/L
3R4F
THS 2.2

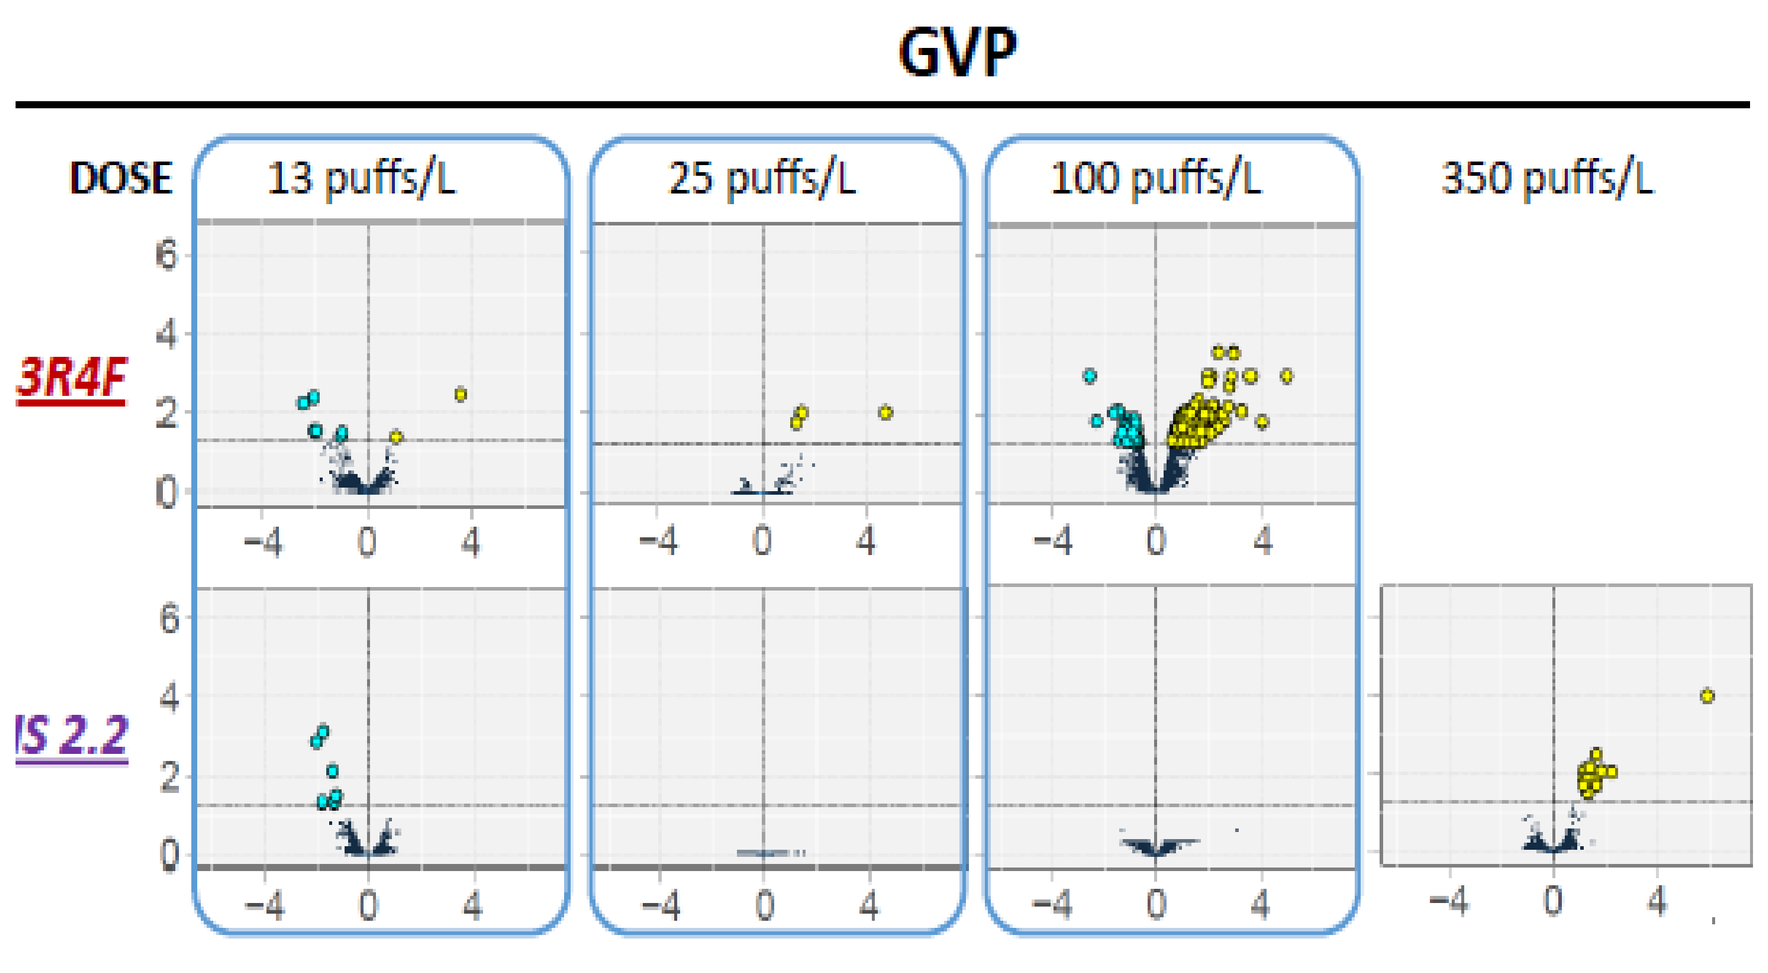

GVP
DOSE
13 puffs/L
25 puffs/L
100 puffs/L
350 puffs/L
3R4F

Figure 2. Gene expression changes in NHBE cells exposed for 4 hours to AE, TPM and GVP fractions generated from 3R4F smoke and THS 2.2 aerosol. For each gene, the gene expression change was calculated as the Log2 FC and the statistical significance as –log10(fdr). The log2 FC are shown on the x axis. The –log10(fdr) are shown in the y axis. Negative fold-changes are shown in the volcano plots in cyan and positive fold-changes in yellow. Changes below a fdr of 0.05 are shown as dark dots.

CONCLUSIONS

- Chemical characterization of mainstream THS2.2 aerosol showed similar nicotine levels and substantially reduced levels of 53 HPHCs compared to mainstream 3R4F smoke.
- Exposure to THS 2.2 aerosol resulted in increased NHBE cell variability and a lower level of toxicity across all HCS endpoints compared to 3R4F smoke fractions. In addition, THS 2.2 aerosol showed a lower level of biological network perturbation compared to 3R4F smoke.
- Taken together, these results suggest that THS 2.2 aerosol is less toxic than cigarette smoke to NHBE cells and thus, may have the potential to reduce the risk of smoking-related diseases.

REFERENCES

(1) The Health Consequences of Smoking—50 Years of Progress: A Report of the Surgeon General. (2014), (U.S. Department of Health and Human Services, C. f. D. C. a. P., National Center for Chronic Disease Prevention and Health Promotion, Office on Smoking and Health, Ed.), Atlanta.

(2) Modified Risk Tobacco Product Applications: Draft Guidance for Industry. (2012), Food and Drug Administration.

(3) Baker, R. R. (1974) Temperature distribution inside a burning cigarette. Nature, 247, 405-406.

(4) Rodgman, A., Perfetti, T. A. (2013) The chemical components of tobacco and tobacco smoke. 2nd edition. CRC Press, Taylor & Francis Group, Boca Raton.

(5) Harmful and Potentially Harmful Constituents in Tobacco Products and Tobacco Smoke; Established List (2012), pp 20034-20037, US Food and Drug Administration

(6) Adamson, J et al. (2011) Jason Adamson, Linsey E Haswell, Gary Phillips and Marianna D Gaça (2011). In Vitro Models of Chronic Obstructive Pulmonary Disease (COPD).

(7) Gonzalez-Suarez, I et al. (2016) In Vitro Systems Toxicology Assessment of a Candidate Modified Risk Tobacco Product Shows Reduced Toxicity Compared to That of a Conventional Cigarette. Chem Res Tox, 29(1): 3-18.

PMI RESEARCH & DEVELOPMENT

Philip Morris International R&D, Philip Morris Products S.A., Quai Jeanrenaud 5, 2000 Neuchâtel, Switzerland (part of Philip Morris International group of companies) www.pmiscience.com