REDUCED EXPOSURE TO HARMFUL AND POTENTIALLY HARMFUL CONSTITUENTS AFTER FIVE DAYS OF USE OF TOBACCO HEATING SYSTEM 2.2: A COMPARISON WITH CONTINUED COMBUSTIBLE CIGARETTE USE OR SMOKING ABSTINENCE [JAPAN]

C. Haziza*[1], G. de La Bourdonnaye[1], G. Baker[1], M. Benzimra[1], J. Ancerewicz[1], P. Picavet[1], H. Miura[2], and F. Lüdicke[1]

[1]: Philip Morris International Research & Development, Neuchatel, Switzerland, [2]: Higashi Shinjuku Clinic, Tokyo, Japan

Introduction and Objectives

The Tobacco Heating System (THS) 2.2 was developed to reduce or eliminate the formation of harmful and potentially harmful smoke constituents (HPHCs) in the aerosol through heating and not burning tobacco, while preserving as much as possible the taste, sensory experience, nicotine delivery profile and ritual characteristics of combustible cigarettes (CC).

The study reported here is part of a global clinical program for THS 2.2 and was designed to demonstrate exposure reduction to selected HPHCs when switching from CC to THS 2.2 use in a controlled setting for 5 days, compared to those continuing to smoke CC. Smokers who abstained from smoking were used as a benchmark. The BoExp measured in this study represent 14 HPHCs, considered to provide a representative assessment of human uptake of a variety of toxicants and carcinogens in tobacco-products.

This study also included endpoints to investigate the acceptance of THS 2.2 as a substitute to CC through assessment of subjective effects (questionnaire of smoking urges-Brief [QSU-Brief]; modified cigarette evaluation questionnaire [mCEQ]).

Methods

- Open-label, randomized, controlled, 3-arm parallel groups, confinement study.
- 160 healthy smokers aged between 23 and65 years.
- Subjects smoked CC during 2 baseline days prior to being randomized for 5 days to the following arms: *ad libitum* CC use; *ad libitum* THS 2.2 use; or smoking abstinence (SA).
- The BoExp were selected based on a variety of criteria:
 - specificity to the source of exposure with other sources being minor or non-existent;
 - detectability using validated methods;
 - reflecting a specific toxic exposure ;
 - representing assessment of both gas and particulate phase of the THS 2.2 aerosol;
 - covering a broad variety of chemical and organ toxicity classes (carcinogen, cardiovascular toxicant, respiratory toxicant, reproductive and development toxicant, addiction potential).
- Urinary BoExp 24h-urine samples were collected daily.
- An analysis of variance (ANOVA), adjusted for log-transformed baseline values, sex and daily CC consumption was applied to the log-transformed BoExp levels with the study arm as a factor.
- The study was conducted in Japan in 2013 according to ICH GCP, approved by an International Review Board, and registered at ClinicalTrials.gov (NCT01970982).

Results

Demographics

Variable	Statistic	THS 2.2 (N=80)	CC (N=40)	SA (N=40)	Overall (N=160)
Age (years)	Mean (SD)	37.6 (11.7)	37.2 (11.7)	35.9 (10.6)	37.1 (11.4)
	Min, Max	23, 64	23, 65	23, 64	23, 65
Sex (Male)	n (%)	40 (50.0%)	20 (50.0%)	20 (50.0%)	80 (50.0%)
Daily CC consumption at screening					
10-19 cig/day	n (%)	44 (55.0%)	22 (55.0%)	21 (52.5%)	87 (54.4%)
> 19 cig/day	n (%)	36 (45.0%)	18 (45.0%)	19 (47.5%)	73 (45.6%)

Biomarkers of Exposure

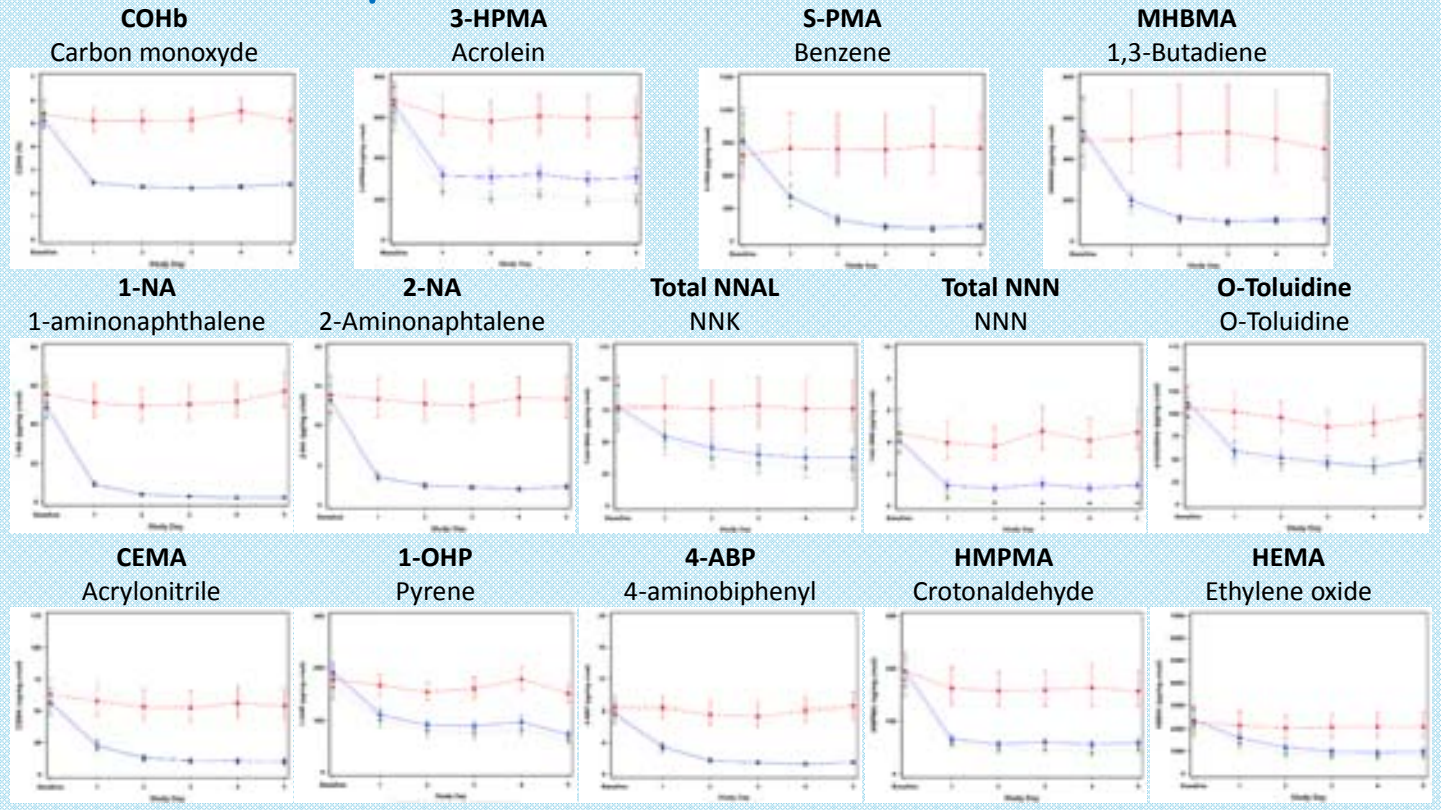
COHb
Carbon monoxyde
3-HPMA
Acrolein
S-PMA
Benzene
MHBMA
1,3-Butadiene
1-NA
1-aminonaphthalene
2-NA
2-Aminonaphtalene
Total NNAL
NNK
Total NNN
NNN
O-Toluidine
O-Toluidine
CEMA
Acrylonitrile
1-OHP
Pyrene
4-ABP
4-aminobiphenyl
HMPMA
Crotonaldehyde
HEMA
Ethylene oxide

Fig 1: Change of Primary BoExp versus CC and SA. Geometric Mean and 95% CI

THS 2.2 — CC — SA

- The reductions in BoExp levels were observed within 24 hours of initiating THS 2.2 use.
- After the 5-day exposure period, the levels of BoExp were significantly reduced by 56% to 98% in the THS 2.2 arm as compared to CC and approaching levels seen in the SA arm. Exposure to toluene via the measurement of S-BMA is not presented, as the levels were similar in all study arms.

Daily Product Use and Nicotine Exposure

Daily Product Use

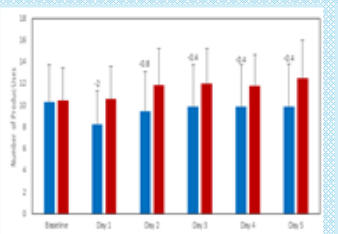

Nicotine Exposure

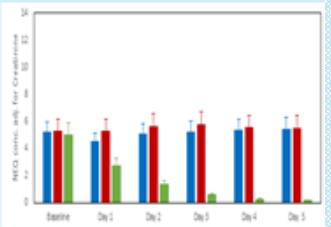

- The average daily product (mean; 95% CI) use between baseline and the end of exposure remained in the same range and was similar in the THS 2.2 and CC arms.
- The total nicotine exposure (mean; 95% CI) measured as nicotine equivalents was also similar in both arms throughout the exposure period (THS 2.2/CC ratio >99% [95% CI: 83% -119%]).

Subjective Effects

Product Evaluation

mCEQ

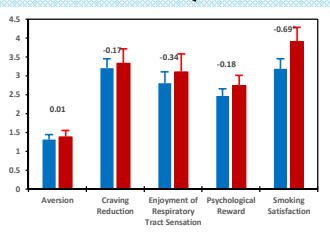
0.01
-0.17
-0.34
-0.18
-0.69
Aversion
Craving Reduction
Enjoyment of Respiratory Tract Sensation
Psychological Reward
Smoking Satisfaction

Urges to Smoke

QSU-Brief – Total Score

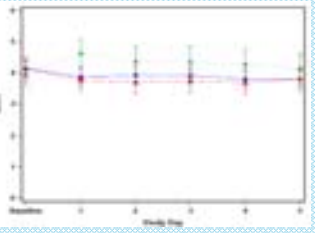

THS 2.2 — CC — SA

- mCEQ subscale scores indicated that THS 2.2 was slightly less satisfying than CC.
- QSU-Brief total score (mean and 95% CI) indicated an equally efficient craving reduction between THS 2.2 and CC over the study period.

Safety

No serious or severe adverse events (AE) were reported during this study, A total of 11 AEs were reported in 10 study subjects in the safety population. All of the AEs were of mild severity. The incidence and frequency of AEs were comparable in the THS 2.2, CC and SA arms.

Conclusions

- The study demonstrated that switching from CC smoking to THS 2.2 use resulted in substantial reductions in exposure to 14 selected HPHCs. The kinetics and the magnitude of decrease of the BoExp levels observed in the THS 2.2 arm were approaching the levels observed in the SA arm.
- The exposure to nicotine was similar between the THS 2.2 and CC arms indicating that users adapted quickly to the new product and achieve their individual nicotine levels.
- These results indicate that THS 2.2 may offer urge to smoke reduction comparable to CC, and could be an acceptable substitute for CC.

ABBREVIATIONS

1-NA: 1-aminonaphtalene; 1-OHP: 1-hydroxypyrene; 2-NA: 2-aminonaphthalene; 3-HPMA: 3-hydroxypropylmercapturic acid; 4-ABP: 4-aminobiphenyl; CEMA: 2-cyanoethylmercapturic acid; COHb: Carboxyhemoglobin; HEMA: 2-hydroxyethyl mercapturic acid; HMPMA: 3-hydroxy-1-methylpropylmercapturic acid; MHBMA: monohydroxybutenyl mercapturic acid; NNAL: 4-(methylnitrosamino)-1-(3-pyridyl)-1-butanol; NNK: 4-(methylnitrosamino)-1-(3-pyridyl)-1-butanone; NNN: N-nitrosonornicotine; S-PMA: S-phenylmercapturic acid

PMI SCIENCE
PHILIP MORRIS INTERNATIONAL

SRNT – 21ST Annual Meeting, Philadelphia, USA
25-28 February 2015

COMPETING FINANCIAL INTERESTS

The research described in this poster was sponsored by Philip Morris International