Proteomics as part of a multi-omics systems toxicological assessment of a mentholated candidate modified risk tobacco product

Nury C.[1], Kogel U.[1], Titz B.[1], Schlage W.K.[2], Martin F.[1], Oviedo A.[3], Lebrun S.[1], Elamin A.[1], Guedj E.[1], Trivedi K.[1], Ivanov N.V.[1], Vanscheeuwijck P.[1], Peitsch M.C.[1], Hoeng J.[1]

[1] PMI R&D, Philip Morris Products S.A., Quai Jeanrenaud 5, CH-2000 Neuchâtel, Switzerland (Part of Philip Morris International group of companies).
[2] Biology consultant, Max-Baermann-Str. 21, 51429 Bergisch Gladbach, Germany.
[3] PMI R&D, Philip Morris International Research Laboratories Pte. Ltd., Science Park II, Singapore (Part of Philip Morris International group of companies)

Introduction and Objectives

Modified risk tobacco products (MRTPs) are being developed with the aim of reducing smoking-related health risks. The Tobacco Heating System 2.2 (THS2.2) is a candidate MRTP that uses the heat-not-burn principle. We engaged a multi-omics systems toxicology approach to assess the respiratory effects of a mentholated THS2.2 (THS2.2M) in a 90-day rat inhalation study (OECD test guideline 413). For this, we complemented the standard OECD-endpoints by quantitative proteomics (iTRAQ) and transcriptomics of nasal epithelium and lung tissue. Quantitative causal network, functional association network, and gene set analyses of the transcriptomics and proteomics data facilitated the identification and comparative assessment of the exposure effects.

The adaptive response of the nasal epithelium to cigarette smoke (CS) included squamous cell metaplasia and an inflammatory response, with high correspondence between the molecular and histopathological results. In contrast to CS exposure, the adaptive tissue and molecular changes to THS2.2M aerosol exposure were much weaker and were limited mostly to the highest THS2.2M concentration in female rats.

In the lung, CS exposure induced an inflammatory response, triggered cellular stress responses, and affected sphingolipid metabolism. These responses were not observed or were much lower after THS2.2M aerosol exposure.

Overall, this multi-omics system toxicology analysis – including quantitative proteomics – complemented and reconfirmed the results from the classical toxicological endpoints as well as identified reductions in a range of additional toxicological pathways [1].

Methods

A

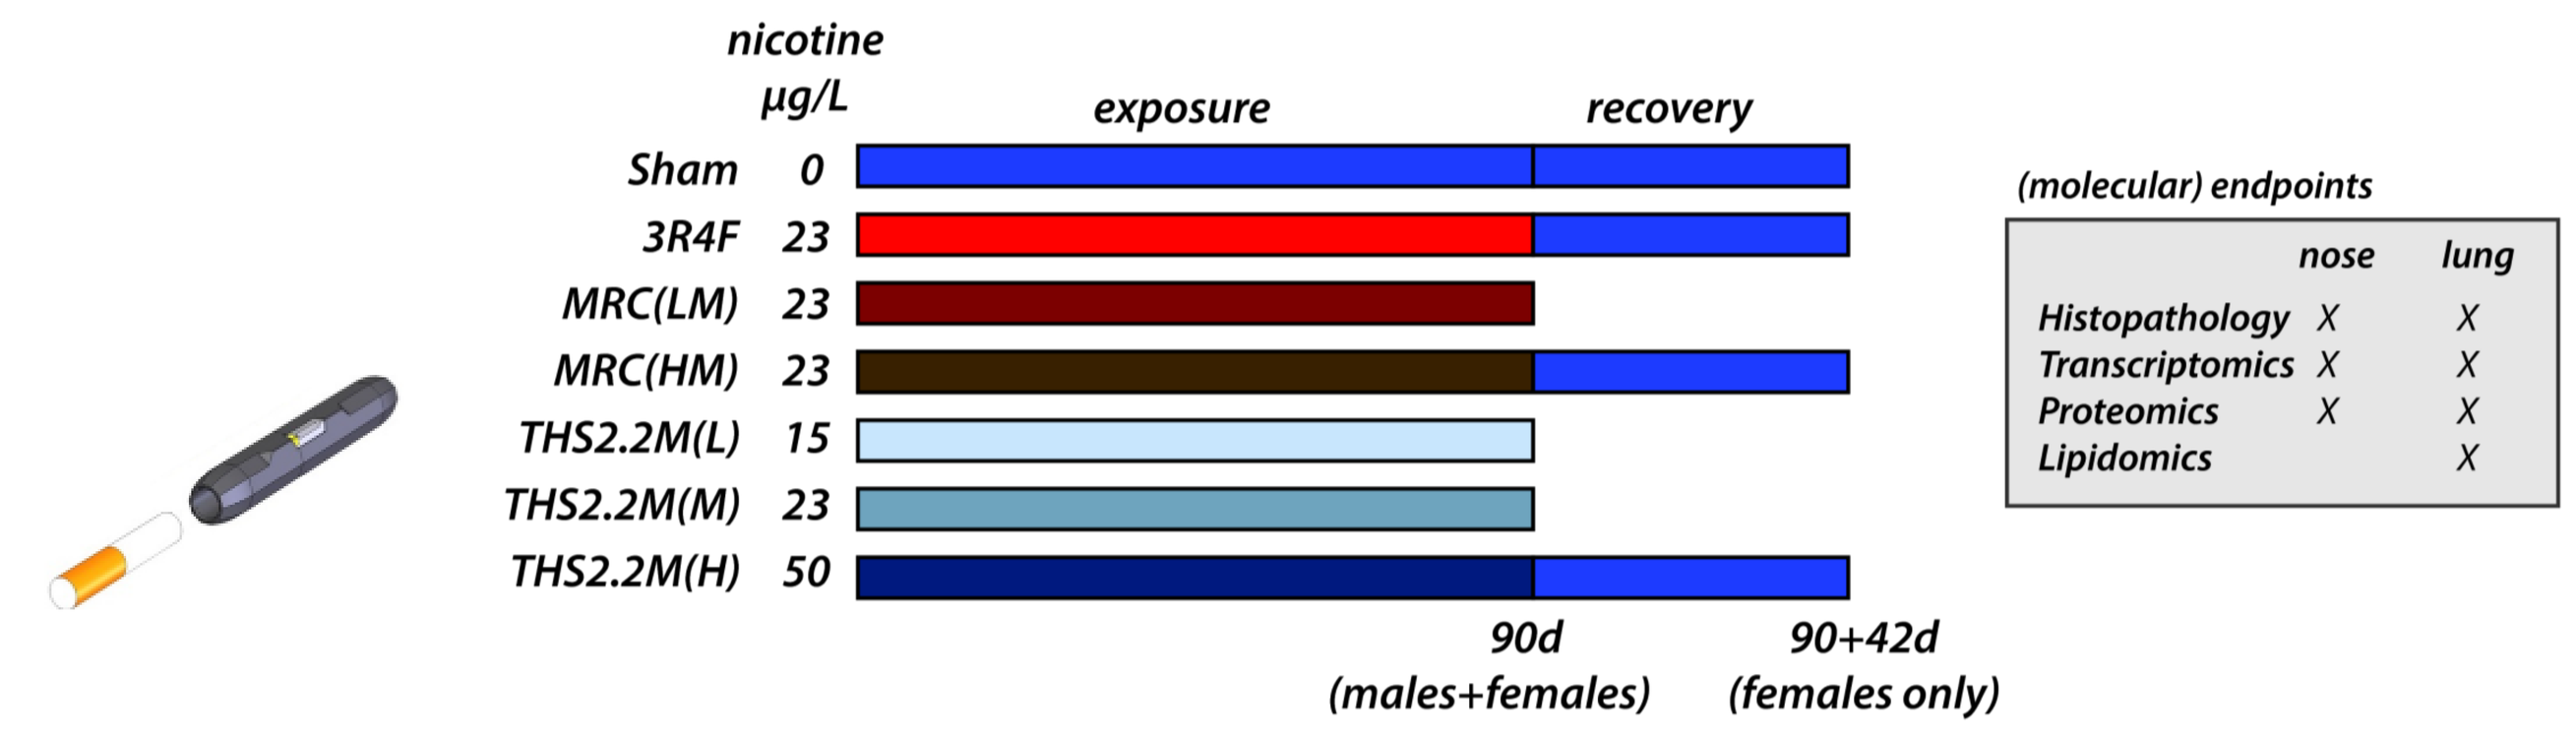
nicotine
µg/L
exposure
recovery
Sham 0
3R4F 23
MRC(LM) 23
MRC(HM) 23
THS2.2M(L) 15
THS2.2M(M) 23
THS2.2M(H) 50
90d
(males+females)
90+42d
(females only)
(molecular) endpoints
nose
lung
Histopathology X X
Transcriptomics X X
Proteomics X X
Lipidomics X

B

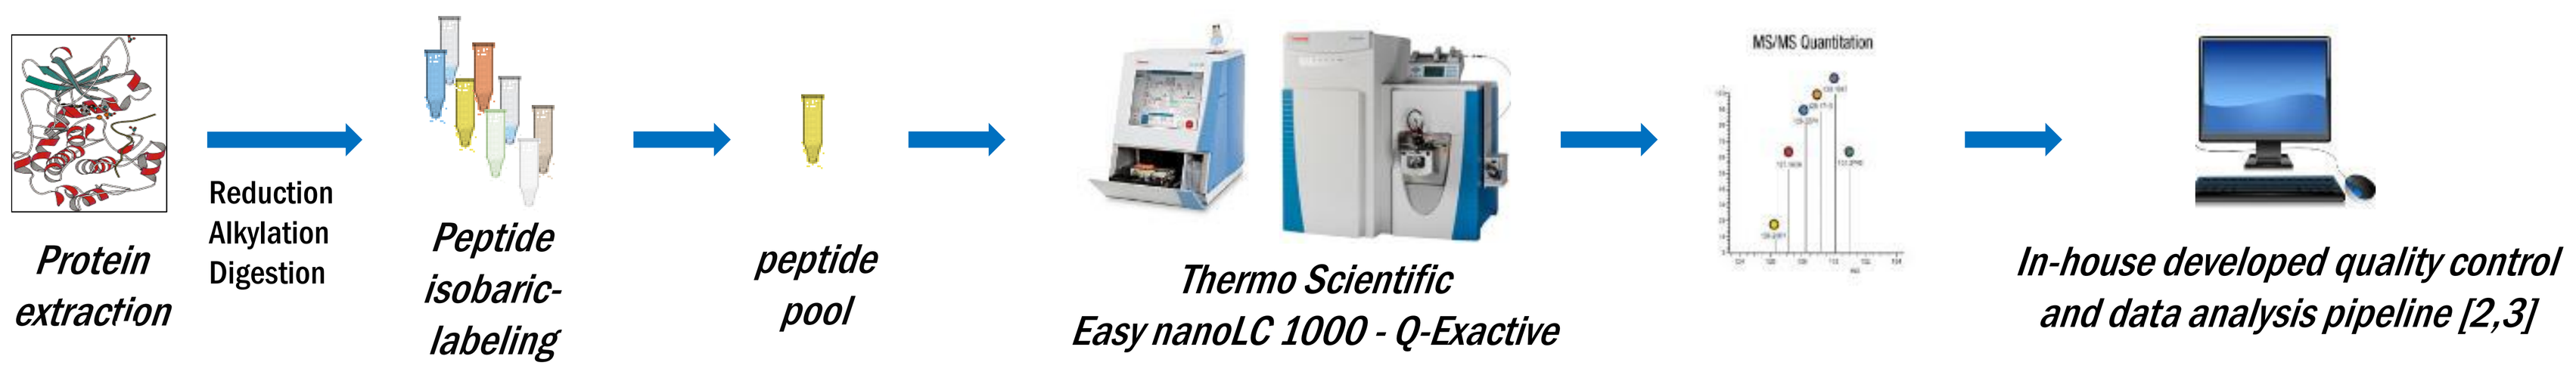
Protein extraction
Reduction
Alkylation
Digestion
Peptide isobaric-labeling
peptide pool
Thermo Scientific
Easy nanoLC 1000 - Q-Exactive
MS/MS Quantitation
In-house developed quality control and data analysis pipeline [2,3]

C

Analysis	samples	each iTRAQ set	#sets	
Set1	all 90d samples	7 sample types + Refmix	12	Sample and channel assignments randomized
Set2	90+42d and corresponding 90d samples	8 sample types	6	

Figure 1. ***Design of the 90-day systems toxicology study to assess effects of CS and THS2.2M exposures on rat respiratory organs.***

A. Groups of male and female rats were exposed for 90 days to fresh air (Sham) or cigarette smoke of three reference cigarettes: a standard reference cigarette (3R4F), and a low menthol (MRC(LM)) and high menthol (MRC(HM)) reference cigarette, all at 23 µg nicotine/L. Groups of rats were exposed to aerosol from THS2.2M (15, 23, and 50 µg/L nicotine). The 90-day exposure period was followed by a 42-day recovery period with female rats exposed to fresh air. Rats were exposed for 6 h per day, for 5 days per week. N=6 for each group.

B. Quantitative proteomics workflow [2,3].

C. Definition of iTRAQ analysis sets.

Results – Lung tissue

A

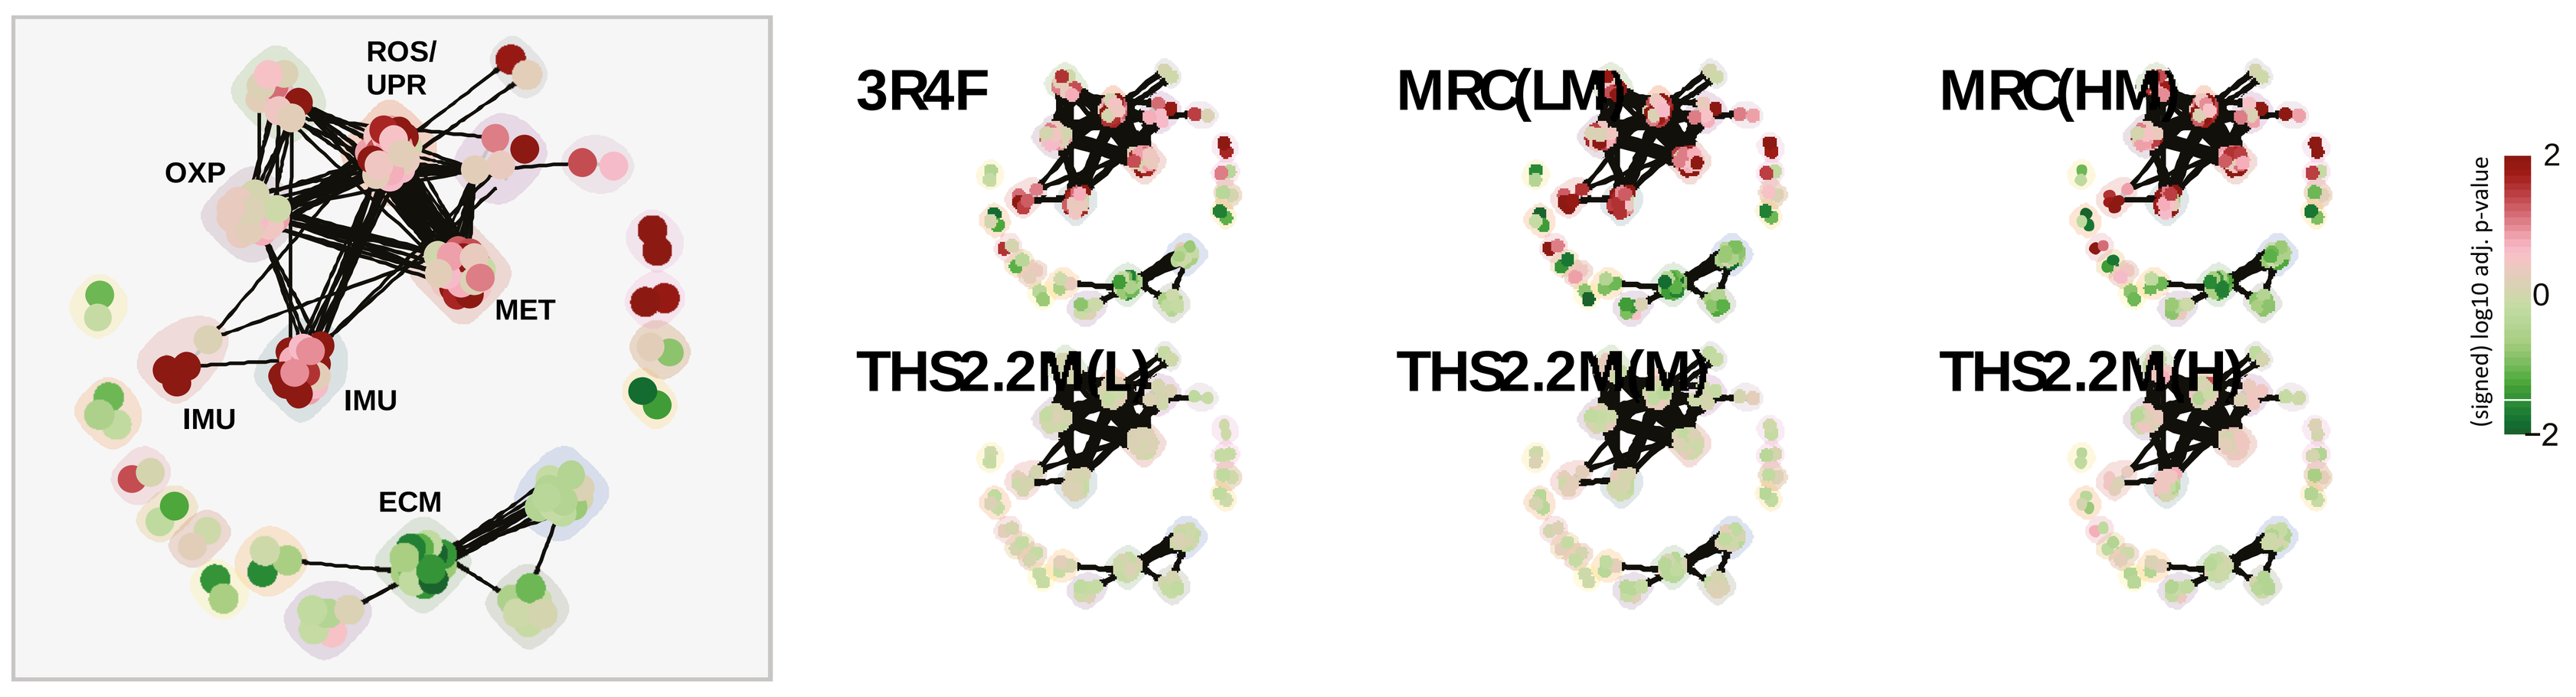
ROS/
UPR
OXP
MET
IMU
IMU
ECM
3R4F
MRC(LM)
MRC(HM)
THS2.2M(L)
THS2.2M(M)
THS2.2M(H)
(signed) log10 adj. p-value

ROS = oxidative-stress response; UPR = unfolded-protein response; OXP = oxidative phosphorylation; MET = metabolism; IMU = immune-related; ECM = extracellular matrix

B

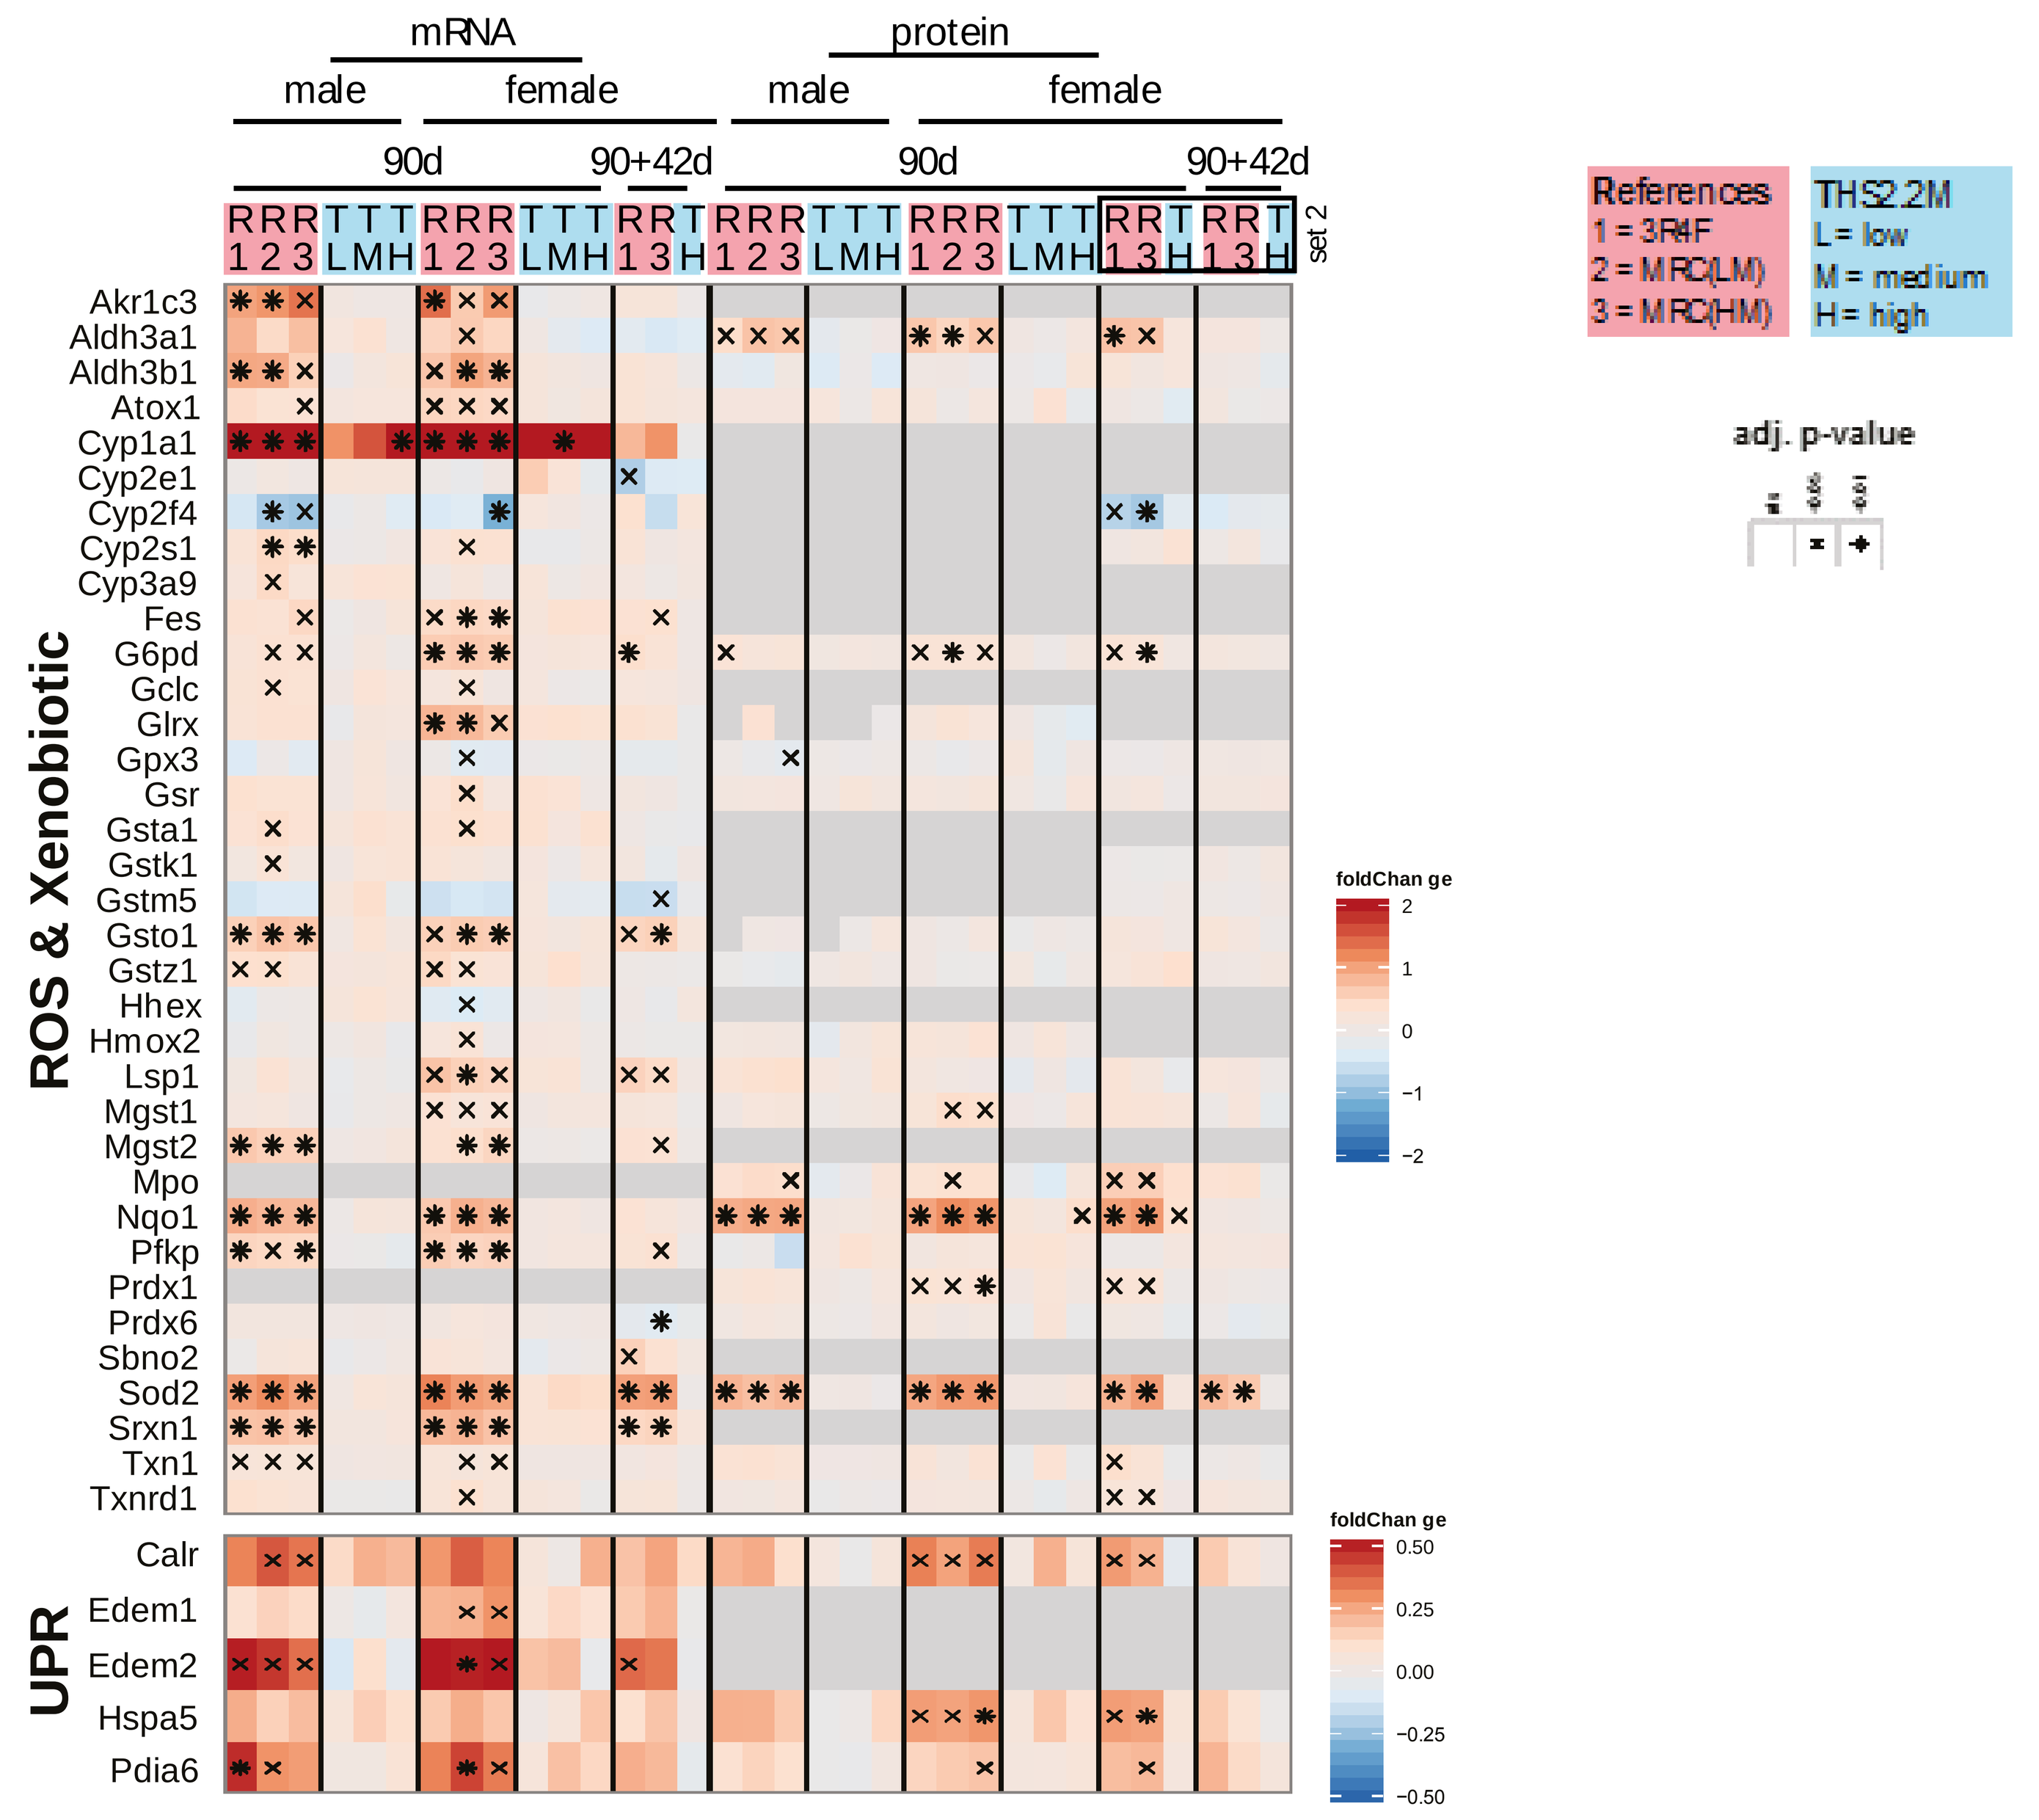
mRNA
protein
male
female
90d
90+42d
set 2
References
1 = 3R4F
2 = MRC(LM)
3 = MRC(HM)
THS2.2M
L = low
M = medium
H = high
adj. p-value
foldChange
ROS & Xenobiotic
UPR
Akr1c3
Aldh3a1
Aldh3b1
Atox1
Cyp1a1
Cyp2e1
Cyp2f4
Cyp2s1
Cyp3a9
Fes
G6pd
Gclc
Glrx
Gpx3
Gsr
Gsta1
Gstk1
Gstm5
Gsto1
Gstz1
Hhex
Hmox2
Lsp1
Mgst1
Mgst2
Mpo
Nqo1
Pfkp
Prdx1
Prdx6
Sbno2
Sod2
Srxn1
Txn1
Txnrd1
Calr
Edem1
Edem2
Hspa5
Pdia6

C

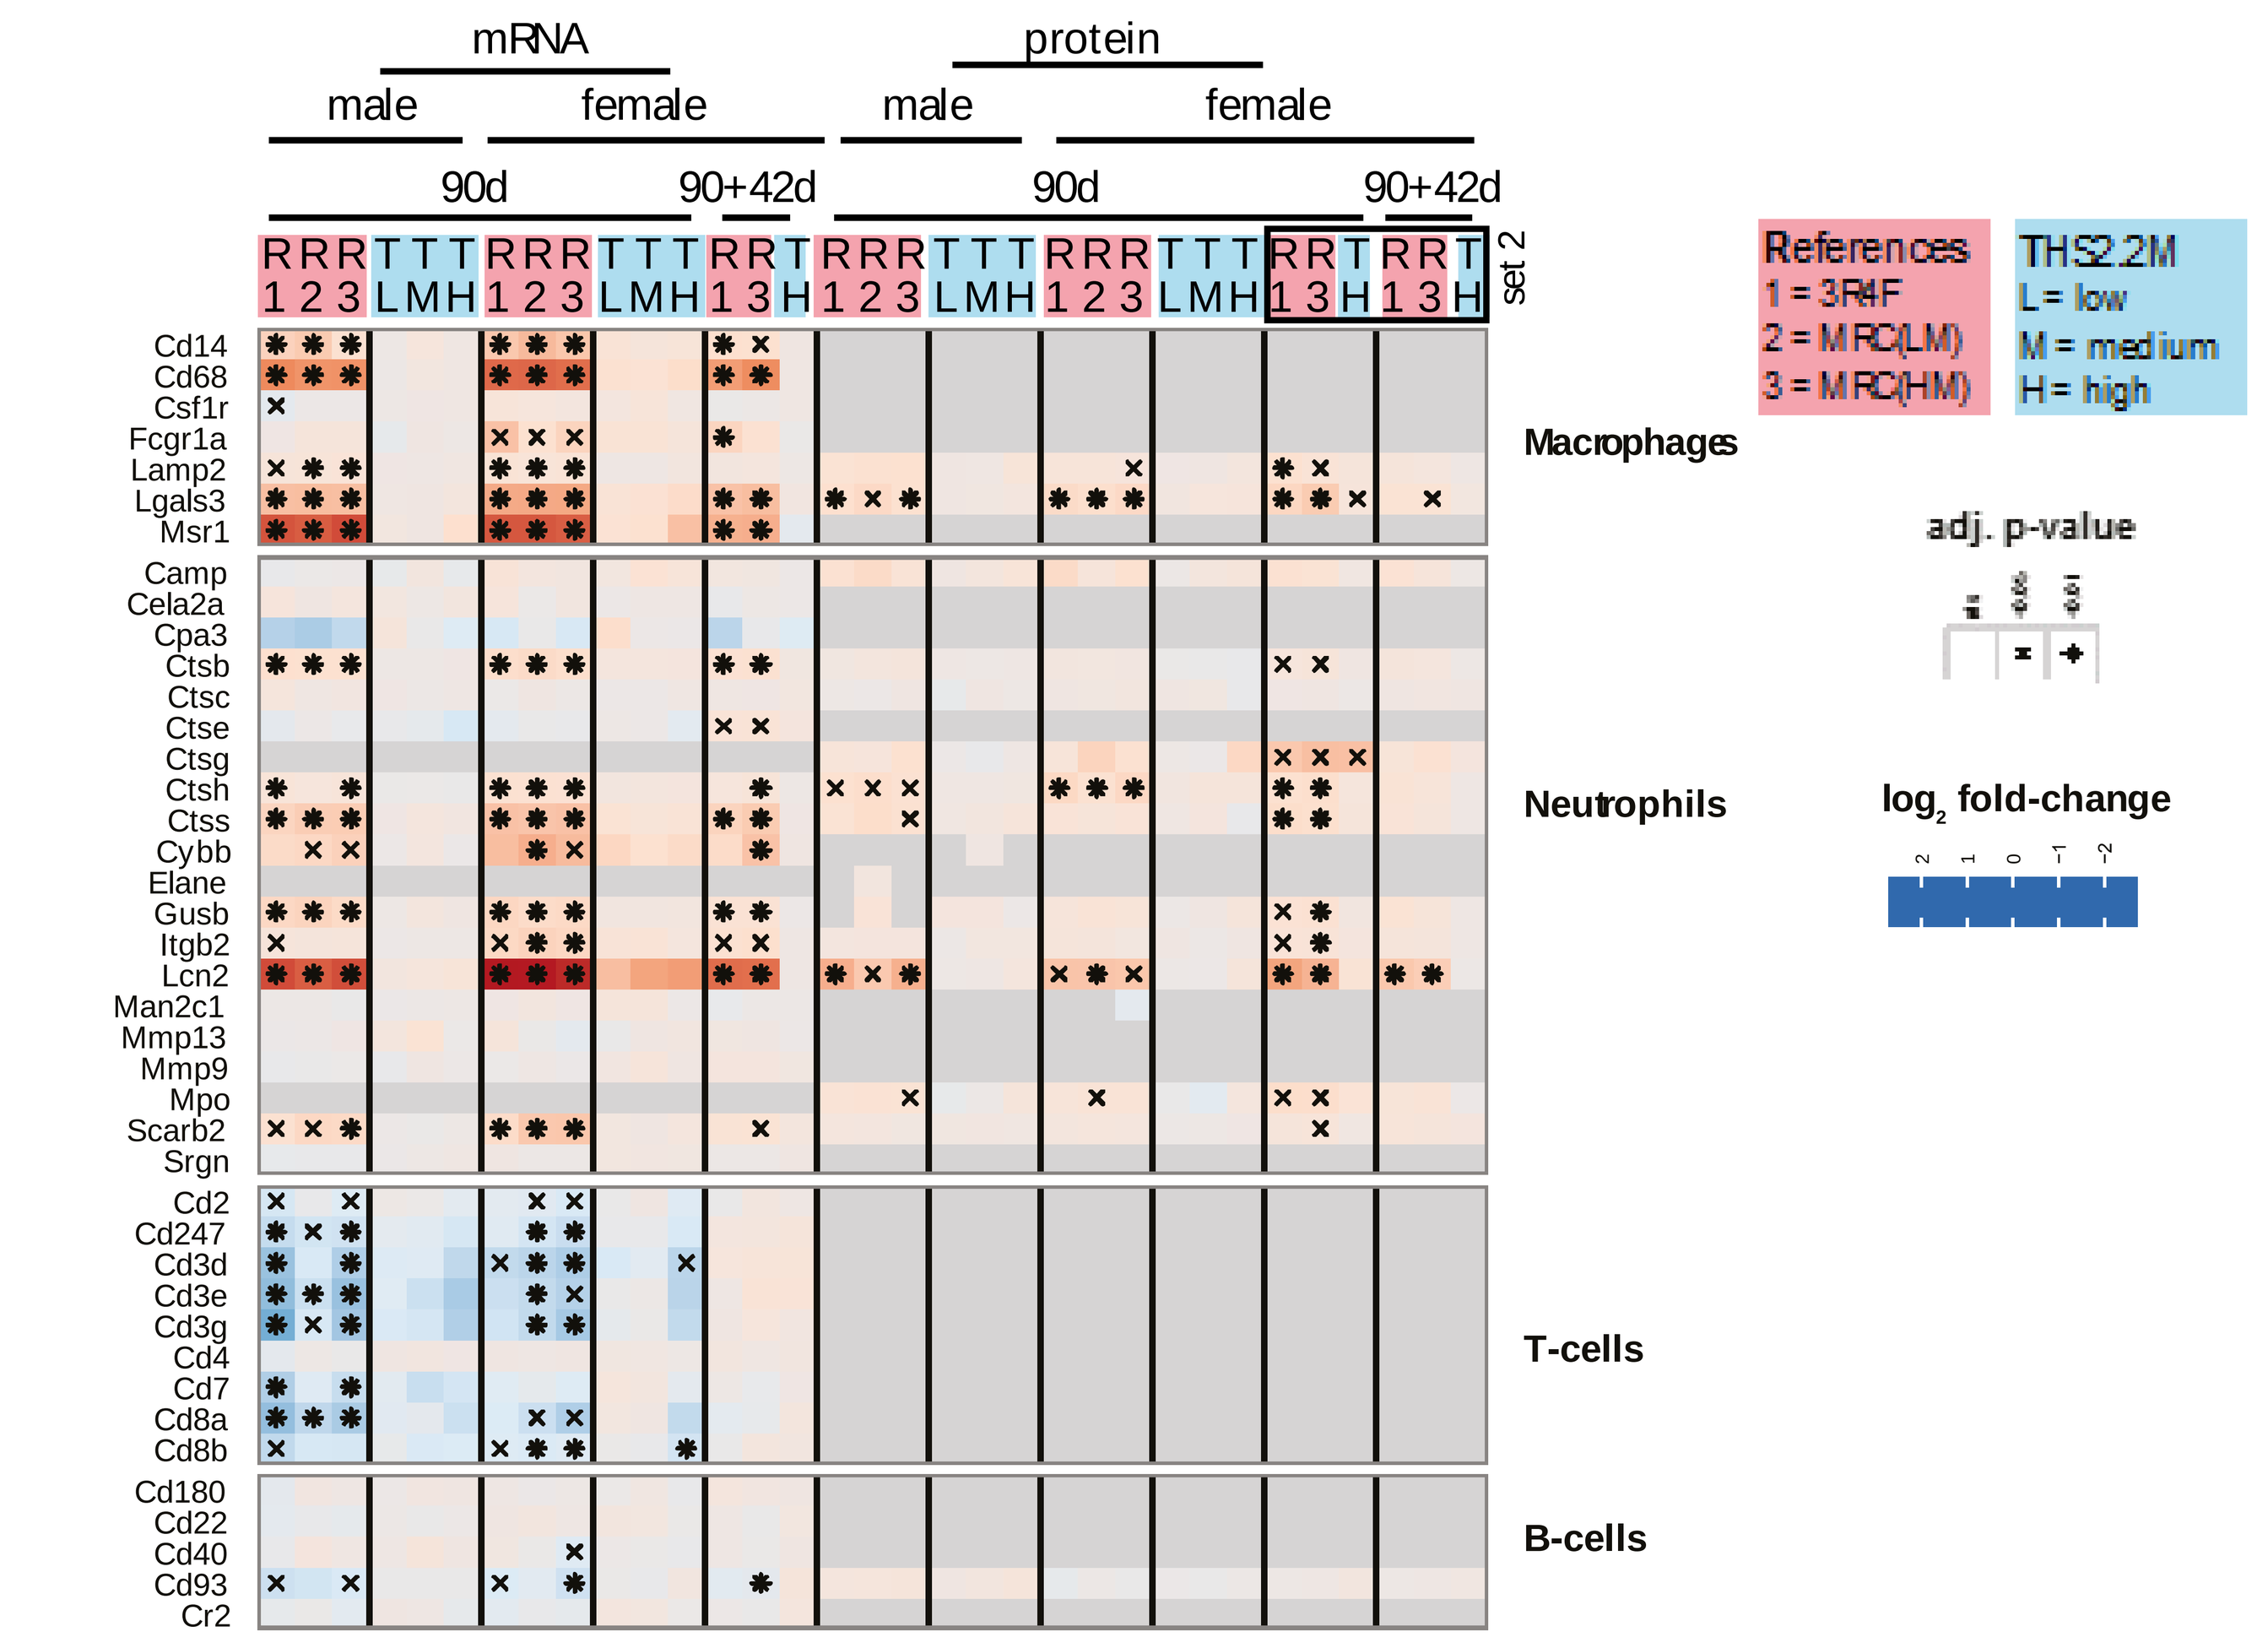
mRNA
protein
male
female
90d
90+42d
set 2
References
1 = 3R4F
2 = MRC(LM)
3 = MRC(HM)
THS2.2M
L = low
M = medium
H = high
adj. p-value
log2 fold-change
Macrophages
Neutrophils
T-cells
B-cells
Cd14
Cd68
Csf1r
Fcgr1a
Lamp2
Lgals3
Msr1
Camp
Cela2a
Cpa3
Ctsb
Ctsc
Ctse
Ctsg
Ctsh
Ctss
Cybb
Elane
Gusb
Itgb2
Lcn2
Man2c1
Mmp13
Mmp9
Mpo
Scarb2
Srgn
Cd2
Cd247
Cd3d
Cd3e
Cd3g
Cd4
Cd7
Cd8a
Cd8b
Cd180
Cd22
Cd40
Cd93
Cr2

Figure 2. ***Response of the lung to exposure.***

A. Functional association clustering for proteomics data of the lung tissue. Functional clusters affected by the exposure conditions were identified and compared across the different clusters, here the 90d female data are shown.

B. Expression/abundance profiles for cellular stress-related gene/protein sets.

C. Expression profiles for panel of immune-cell markers.

Results – Nasal Epithelium

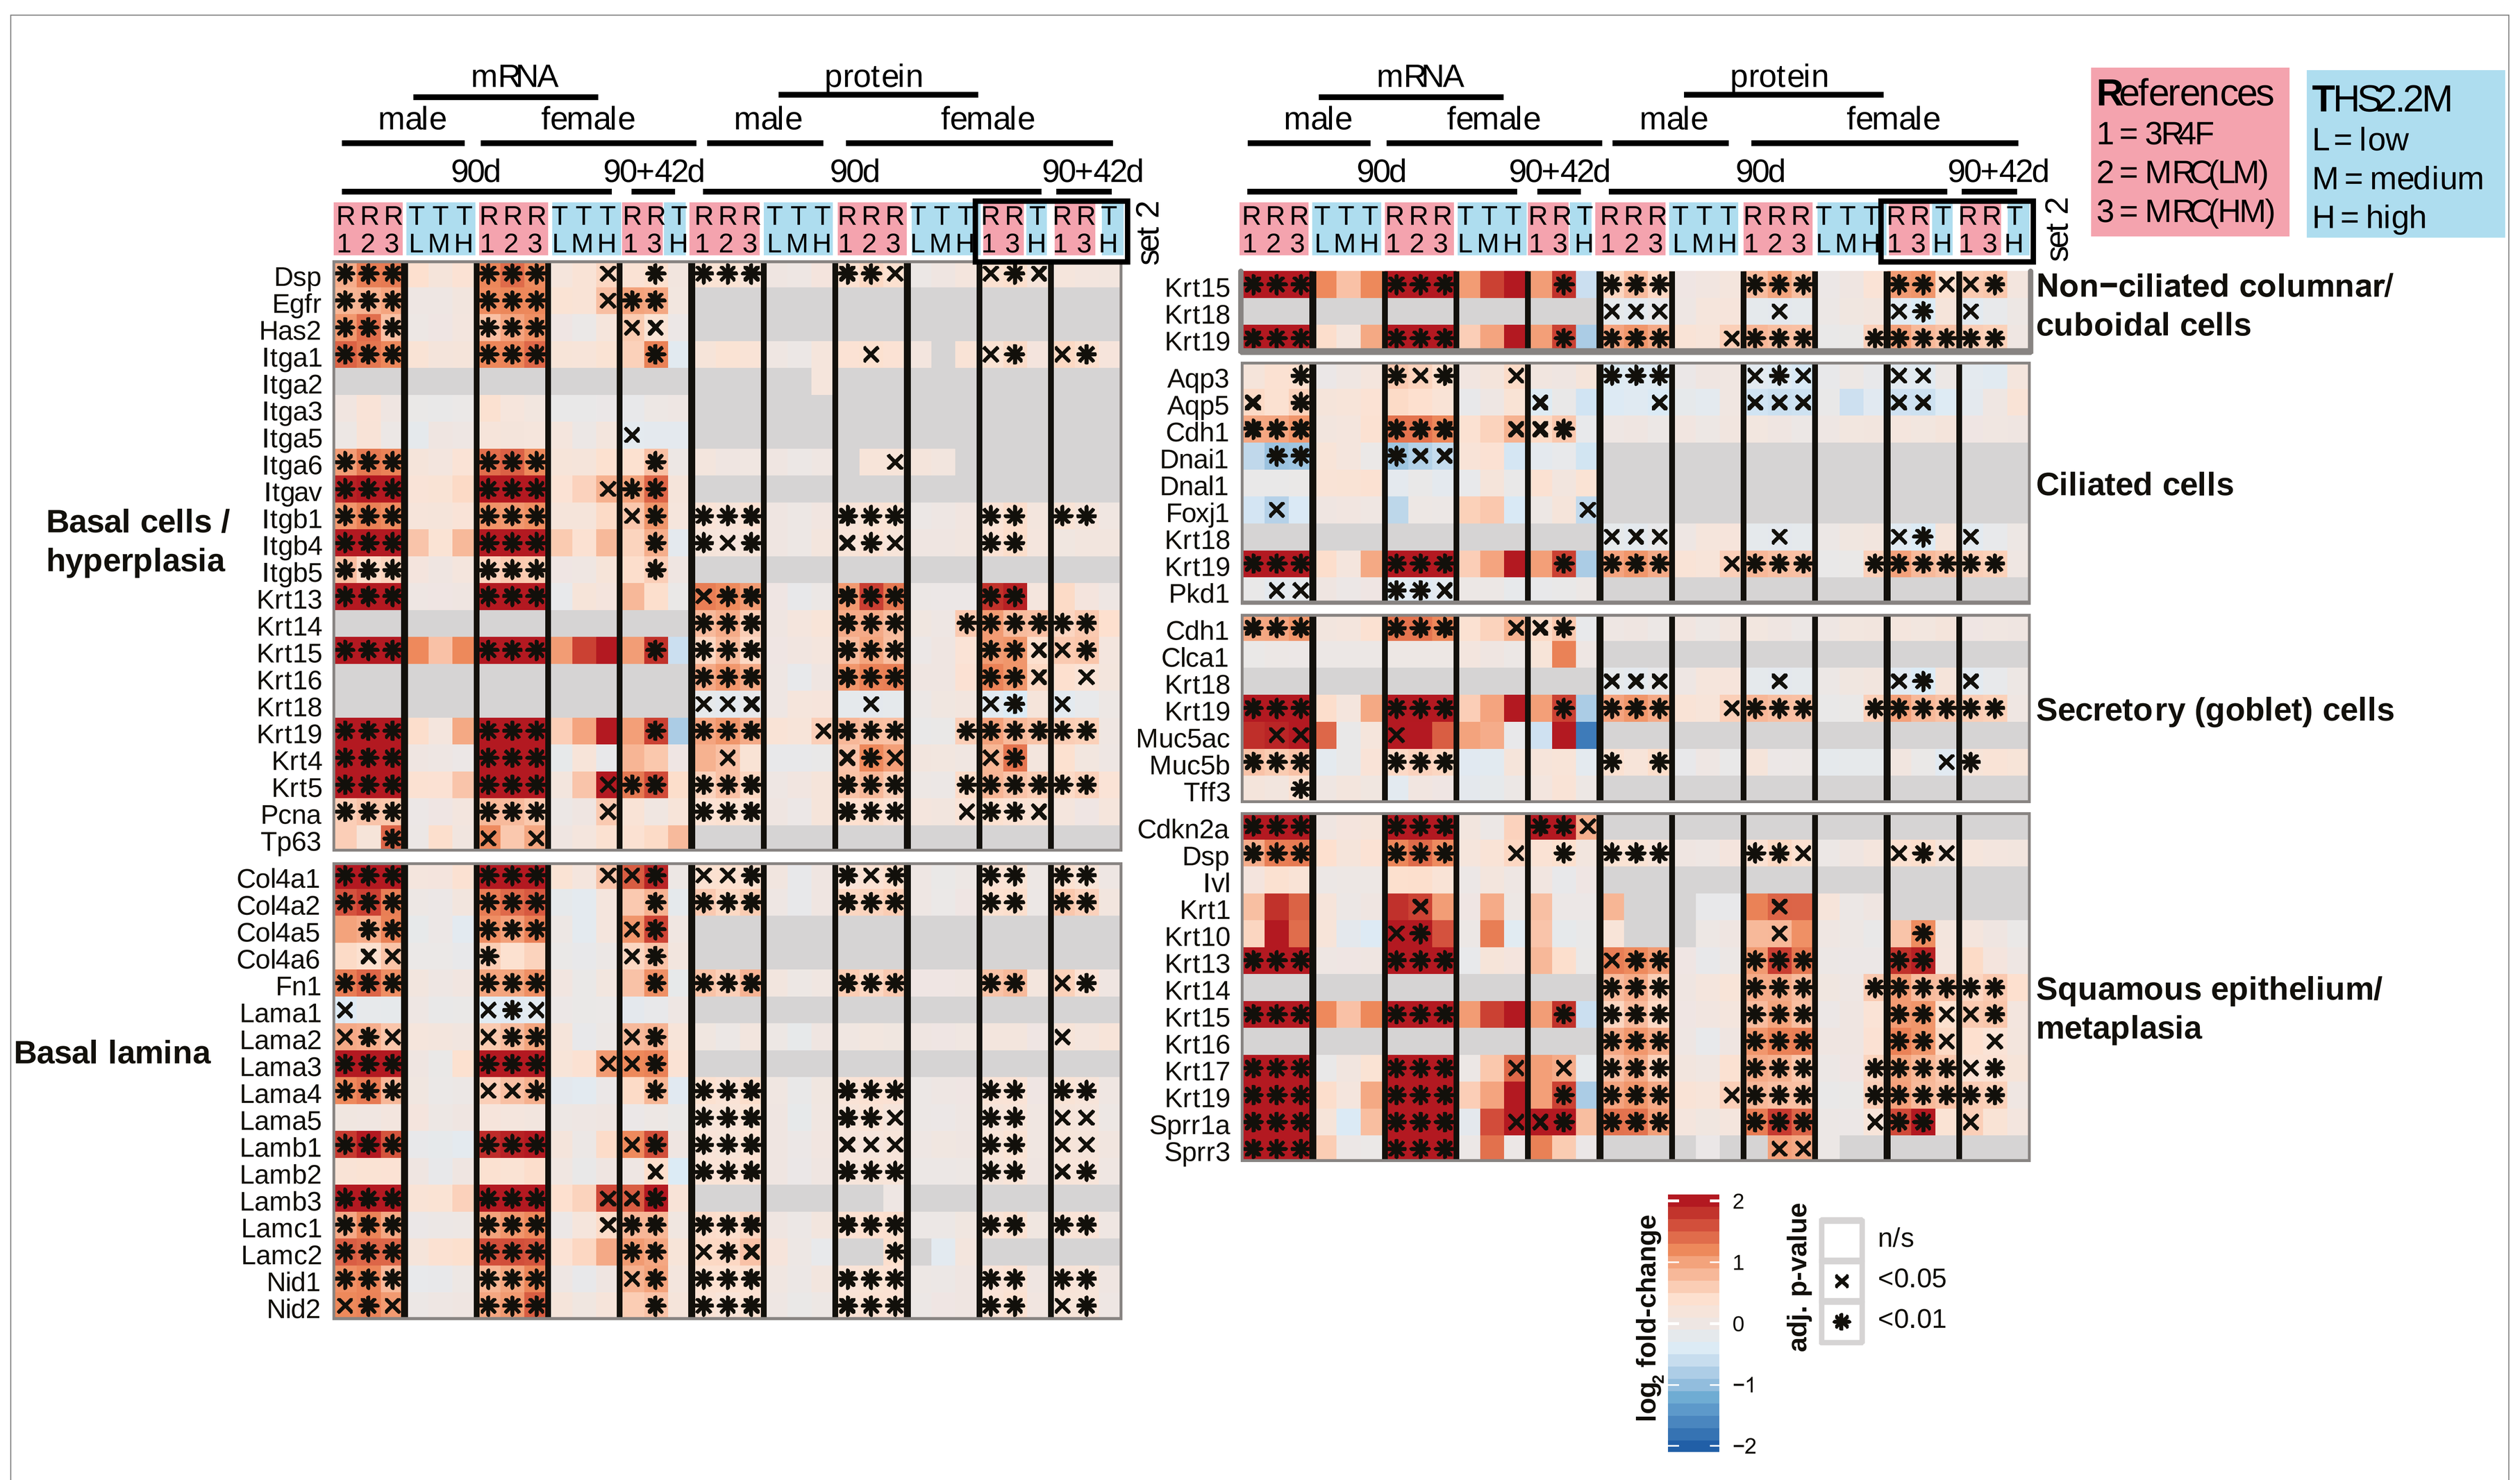
mRNA
protein
male
female
90d
90+42d
set 2
References
1 = 3R4F
2 = MRC(LM)
3 = MRC(HM)
THS2.2M
L = low
M = medium
H = high
Basal cells / hyperplasia
Basal lamina
Non−ciliated columnar/ cuboidal cells
Ciliated cells
Secretory (goblet) cells
Squamous epithelium/ metaplasia
log2 fold-change
adj. p-value
n/s
<0.05
<0.01

Figure 3. ***Response of the respiratory nasal epithelium to exposure.***

*Expression profiles for marker panel of epithelium cell types and basal lamina components. The protein and gene abundance and expression fold-changes compared with the respective Sham groups are color-coded and statistical significance is marked (adjusted p-value, x = < 0.05, * = < 0.01). Missing values are marked in grey.*

Conclusions

- Systems toxicology analysis supported an integrated product risk assessment of THS2.2M. Consistently, the THS2.2M aerosols showed lower effects compared with smoke from reference cigarettes on the molecular profiles and molecular response networks of respiratory nasal epithelium and lung tissue.
- Overall, our systems toxicology endpoints complemented and augmented the more classical endpoints of the OECD TG413 90-day rat inhalation study [4].
- Furthermore, this study revealed activated toxicity pathways and associated candidate biomarkers that could further facilitate the development of new toxicology assessment approaches

References

[1] Kogel, U., et al., Evaluation of the Tobacco Heating System 2.2. Part 7: Systems toxicological assessment of a mentholated version revealed reduced cellular and molecular exposure effects compared with mentholated and non-mentholated cigarette smoke, Regulatory Toxicology and Pharmacology (2016) 81, 123-138.
[2] Titz, B., et al. Proteomics for systems toxicology. Computational and structural biotechnology journal (2014) 11, 73-90.
[3] Titz, B., et al. Analysis of Proteomic Data for Toxicological Applications. Computational Systems Toxicology (2015), Humana Press, pp. 257-284.
[4] Oviedo, A. et al. Evaluation of the Tobacco Heating System 2.2. Part 6: 90-day OECD 413 rat inhalation study with systems toxicology endpoints demonstrates reduced exposure effects of a mentholated version compared with mentholated and non-mentholated cigarette smoke, Regulatory Toxicology and Pharmacology (2016) 81, 93-122.

PMI SCIENCE
PHILIP MORRIS INTERNATIONAL

Human Proteome Organization World Congress Dublin, Ireland

17–21 September 2017

Competing Financial Interest

All authors are employees of Philip Morris International (PMI) or worked for PMI under contractual agreements. PMI is the sole source of funding and sponsor of this project.