Integration of Targeted Proteomics into Systems Toxicology Approach: Candidate Modified-Risk Tobacco Product Assessment using Nasal Cultures

T. Schneider, A. Elamin, A.R. Iskandar, F. Zanetti, C. Merg, A. Sewer, B. Titz, P. Leroy, E. Guedj, S. Majeed, L. Ortega-Torres, W. Schlage, C. Mathis, F. Martin, S. Frentzel, N. V. Ivanov, M.C. Peitsch, J. Hoeng

Philip Morris International R&D, Philip Morris Products S.A., Quai Jeanrenaud 5, 2000 Neuchâtel, Switzerland
(Part of Philip Morris International group of companies)

Introduction and Objectives

Systems biology combines comprehensive molecular analyses and quantitative modeling to understand the characteristics of a biological system as a whole. Leveraging a similar approach, Systems Toxicology aims to decipher complex biological responses following exposures.
This work reports parts of a Systems Toxicology analysis in the context of the *in vitro* assessment of a candidate modified-risk tobacco product (cMRTP) using a human nasal organotypic culture model[1,2]. The term "modified risk tobacco product" means any tobacco product that is sold or distributed for use to reduce harm or risk of tobacco-related diseases associated with commercially marketed tobacco products"[3].
The objectives of this study included to demonstrate (1) robustness and reproducibility of systems biology data obtained from an organotypic *in vitro* nasal culture model for cigarette smoke (CS) and aerosol exposure assessment; (2) relevancy of this approach in a comparative risk assessment of aerosols from a cMRTP compared with CS; and (3) the benefit of including complementary data modalities such as targeted proteomics in this assessment approach.
Complementing a series of functional measures, a causal network enrichment analysis of transcriptomic data was used to compare quantitatively the biological impact of aerosol from the Tobacco Heating System (THS) 2.2, a candidate MRTP, with 3R4F CS at similar nicotine concentrations. In addition, the tissue response was measured by a mass-spectrometry based targeted proteomics approach, parallel reaction monitoring (PRM), and antibody-based multi-analyte profiling (MAP).

Methods

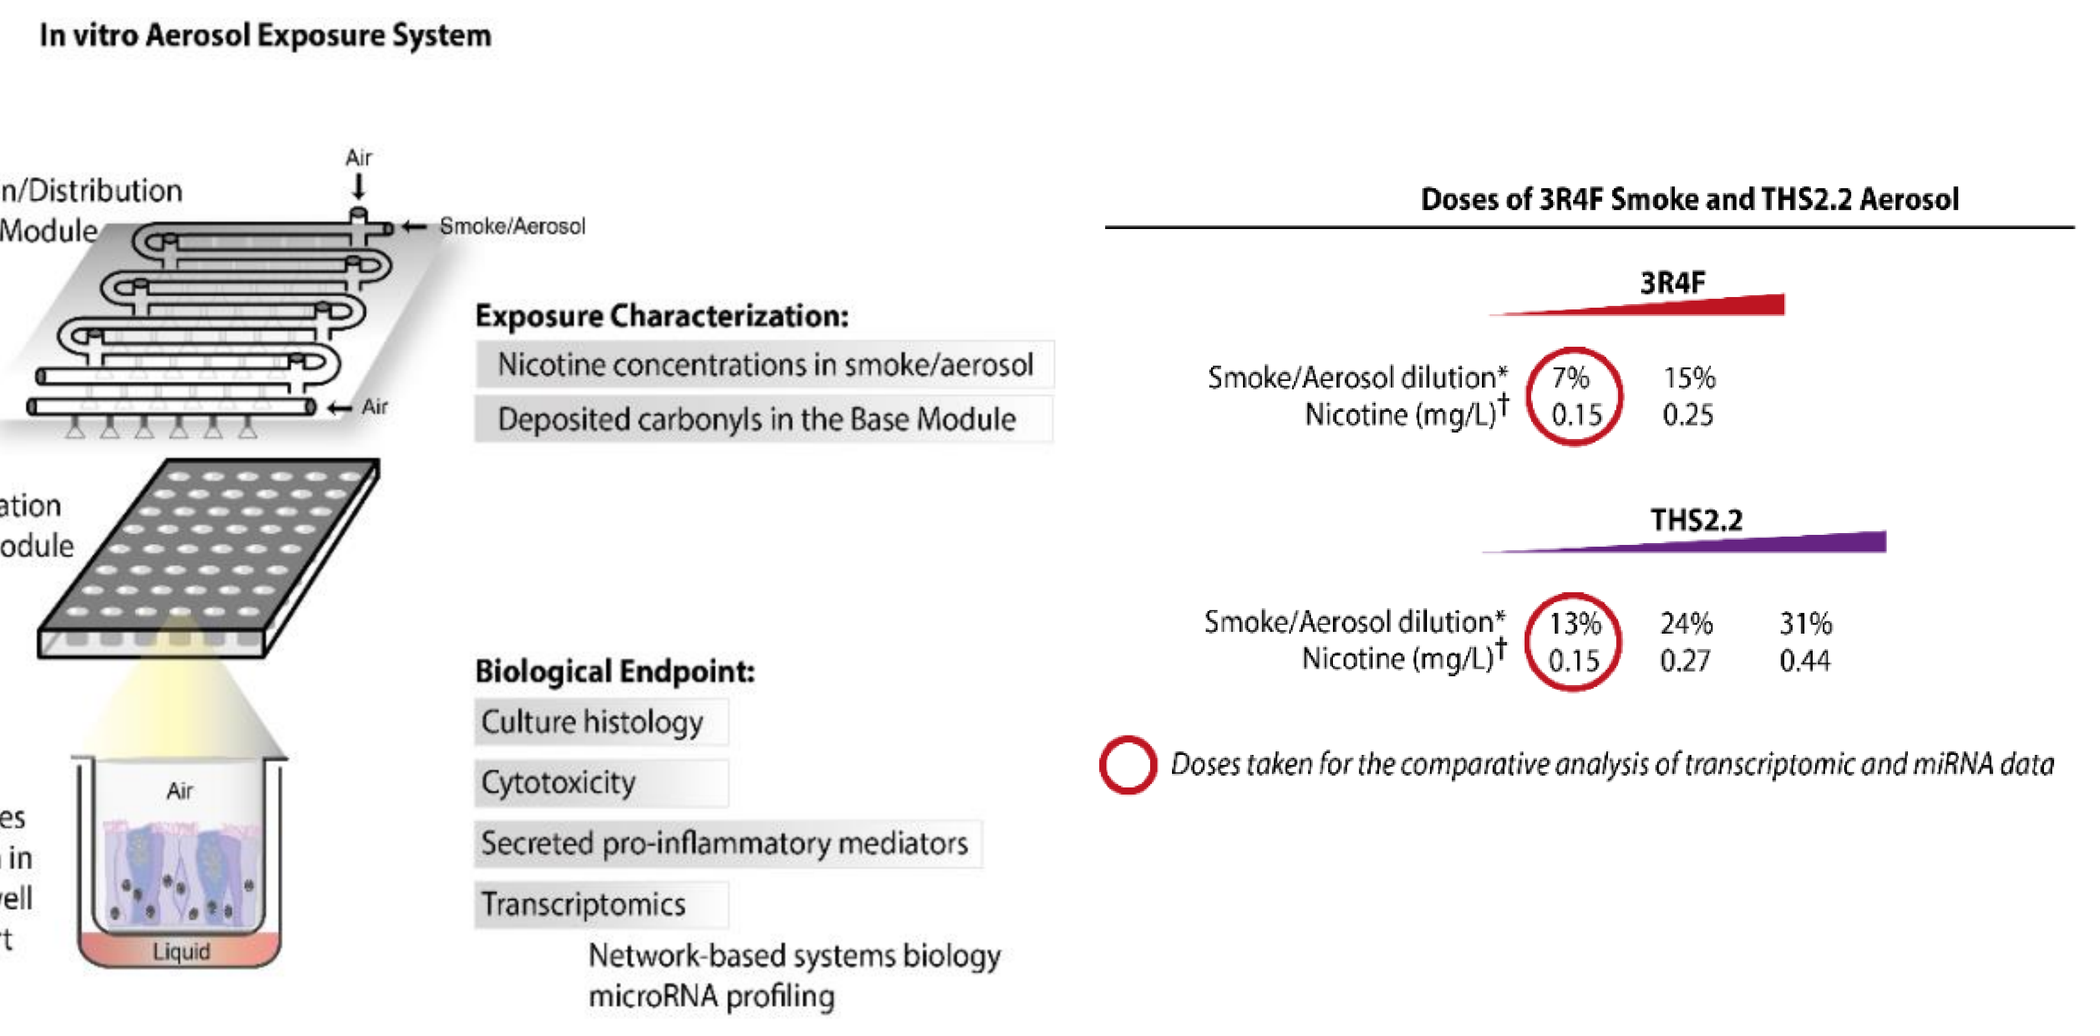
In vitro Aerosol Exposure System
Dilution/Distribution Module
Air
Smoke/Aerosol
Cultivation Base Module
Cultures grown in Transwell Insert
Liquid
Exposure Characterization:
Nicotine concentrations in smoke/aerosol
Deposited carbonyls in the Base Module
Biological Endpoint:
Culture histology
Cytotoxicity
Secreted pro-inflammatory mediators
Transcriptomics
Network-based systems biology
microRNA profiling
Doses of 3R4F Smoke and THS2.2 Aerosol
3R4F
Smoke/Aerosol dilution* 7% 15%
Nicotine (mg/L)† 0.15 0.25
THS2.2
Smoke/Aerosol dilution* 13% 24% 31%
Nicotine (mg/L)† 0.15 0.27 0.44
Doses taken for the comparative analysis of transcriptomic and miRNA data

Figure 1. Study overview
*An organotypic nasal culture model was exposed (acute, for 28 min) to 3R4F CS or THS2.2 aerosol at similar nicotine concentrations in an Exposure System (Vitrocell 24/48®). * Dilution refers to the percent 3R4F smoke or THS2.2 aerosol diluted with air in the Dilution/Distribution Module of the Exposure System. † Nicotine concentration (mg/L) refers the corresponding concentration to the specific dilution of smoke/aerosol determined by trapping the diluted smoke/aerosol in the EXtrelut® 3NT column. The nasal organotypic cultures were reconstituted from the primary nasal epithelial cells of 30 year-old non-smoker male. Figure adapted from Iskandar et al. (2017)[1].*

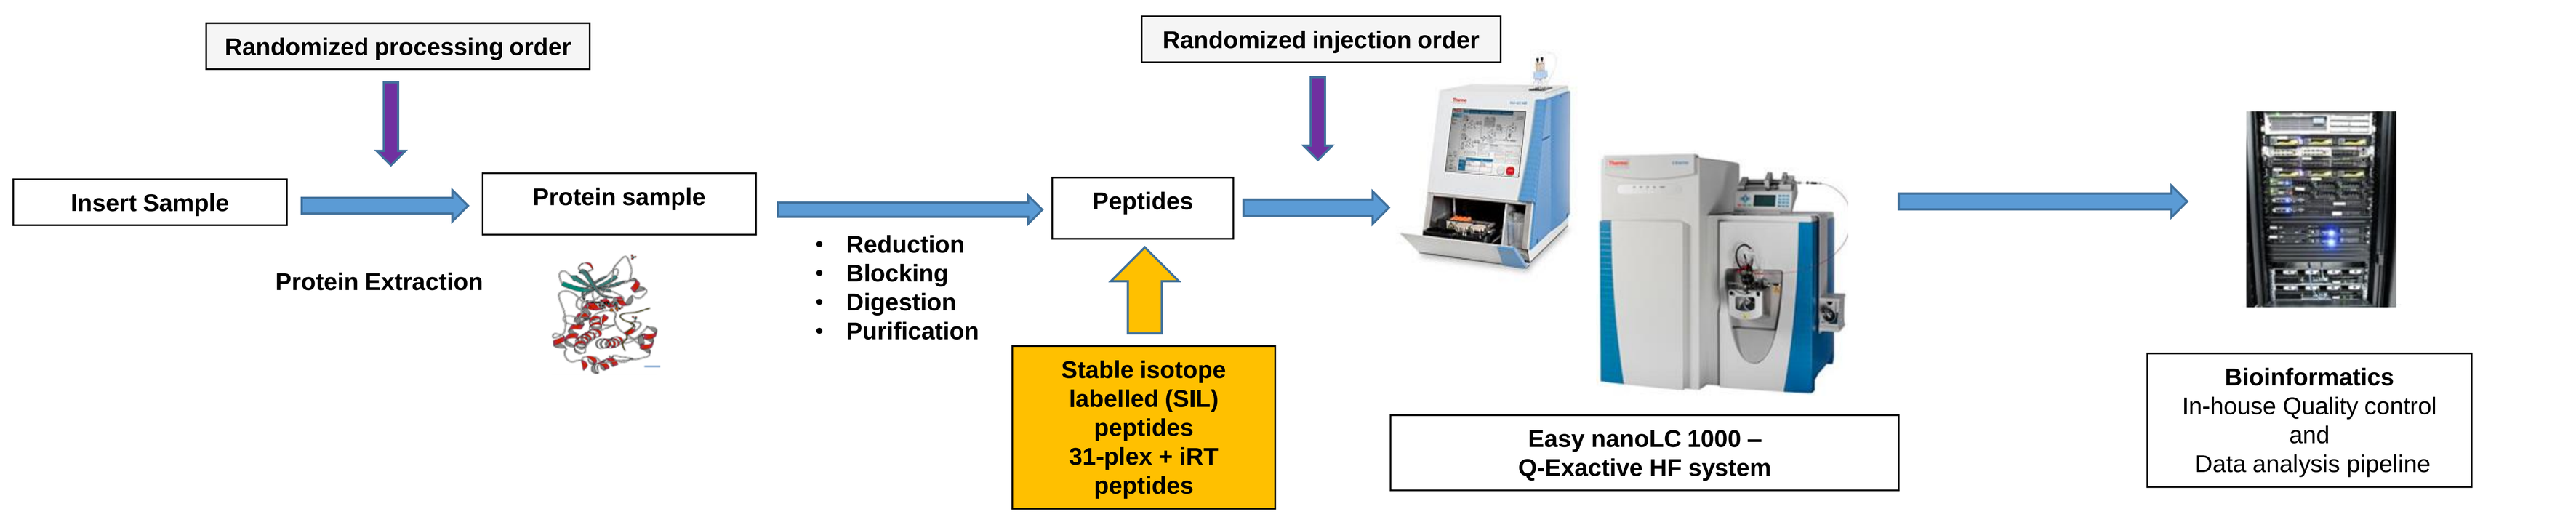
Randomized processing order
Randomized injection order
Insert Sample
Protein sample
Protein Extraction
Reduction
Blocking
Digestion
Purification
Peptides
Stable isotope labelled (SIL) peptides 31-plex + iRT peptides
Easy nanoLC 1000 – Q-Exactive HF system
Bioinformatics
In-house Quality control and Data analysis pipeline

Figure 2. Schematic overview of the parallel reaction monitoring (PRM) workflow. *Absolute quantification of target proteins was achieved by spiking stable isotope labelled (SIL) peptides in known concentration as reference standard. For analysis quality-control and retention time scheduling indexed retention time (iRT)-kit was used[4].*

Cytotoxicity

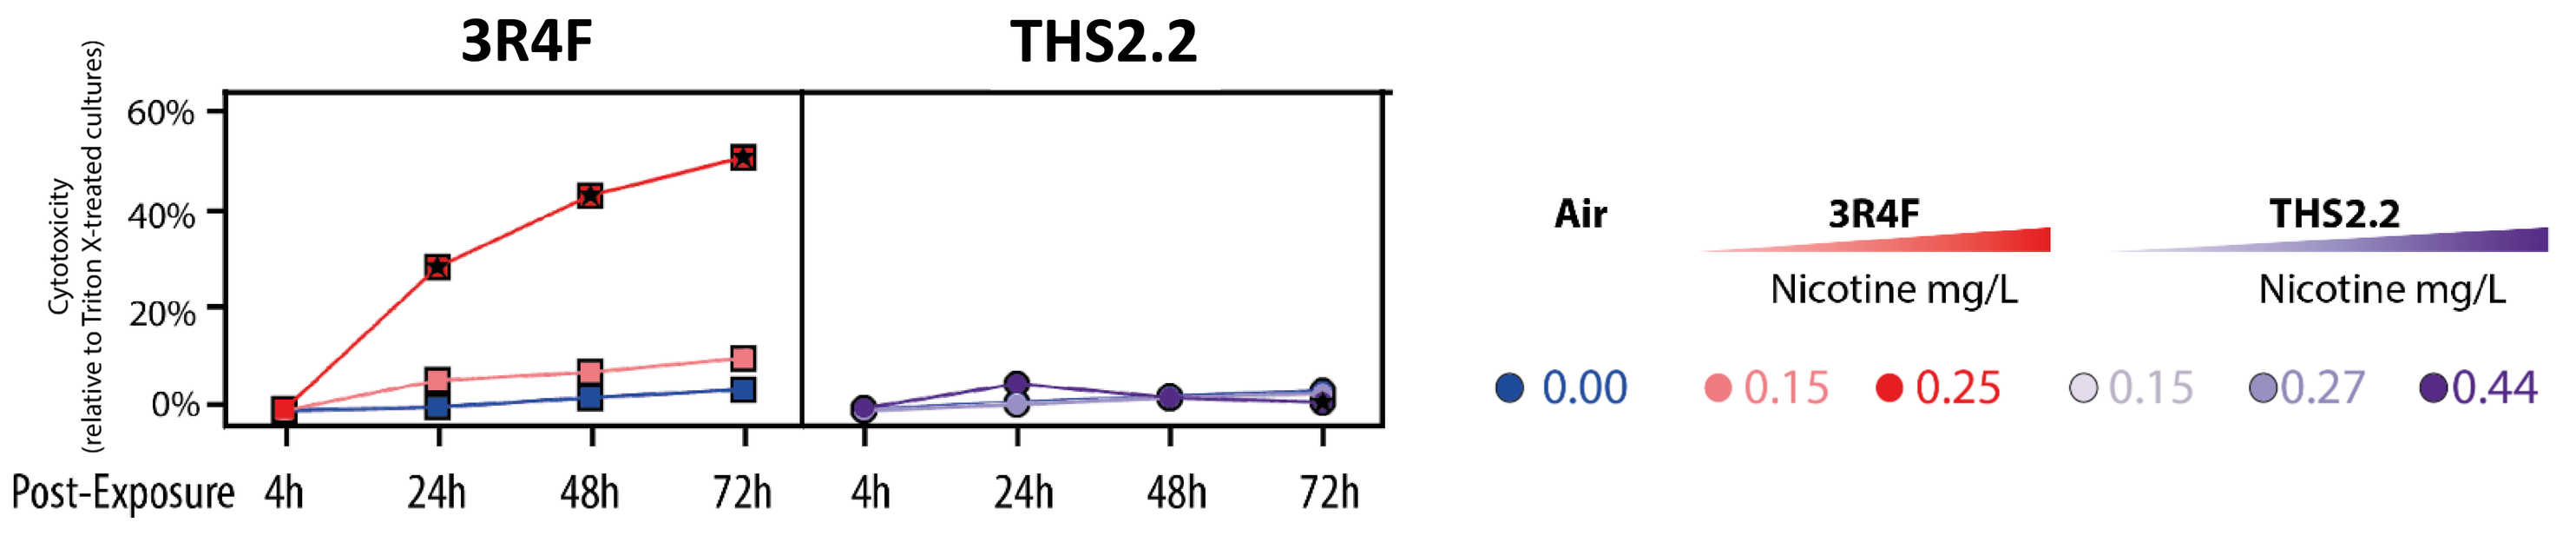
3R4F
THS2.2
Cytotoxicity (relative to Triton X-treated cultures)
60%
40%
20%
0%
Post-Exposure 4h 24h 48h 72h 4h 24h 48h 72h
Air
3R4F Nicotine mg/L
THS2.2 Nicotine mg/L
0.00
0.15 0.25
0.15 0.27 0.44

Figure 3. Assessment of cytotoxicity. *Cytotoxicity levels following exposure were measured based on the levels of adenylate kinase activity in the basolateral media (adenylate kinase release assay) at various post-exposure time points in the nasal cultures.*

Transcriptomics and Network-Based Analysis of the Exposure Impact

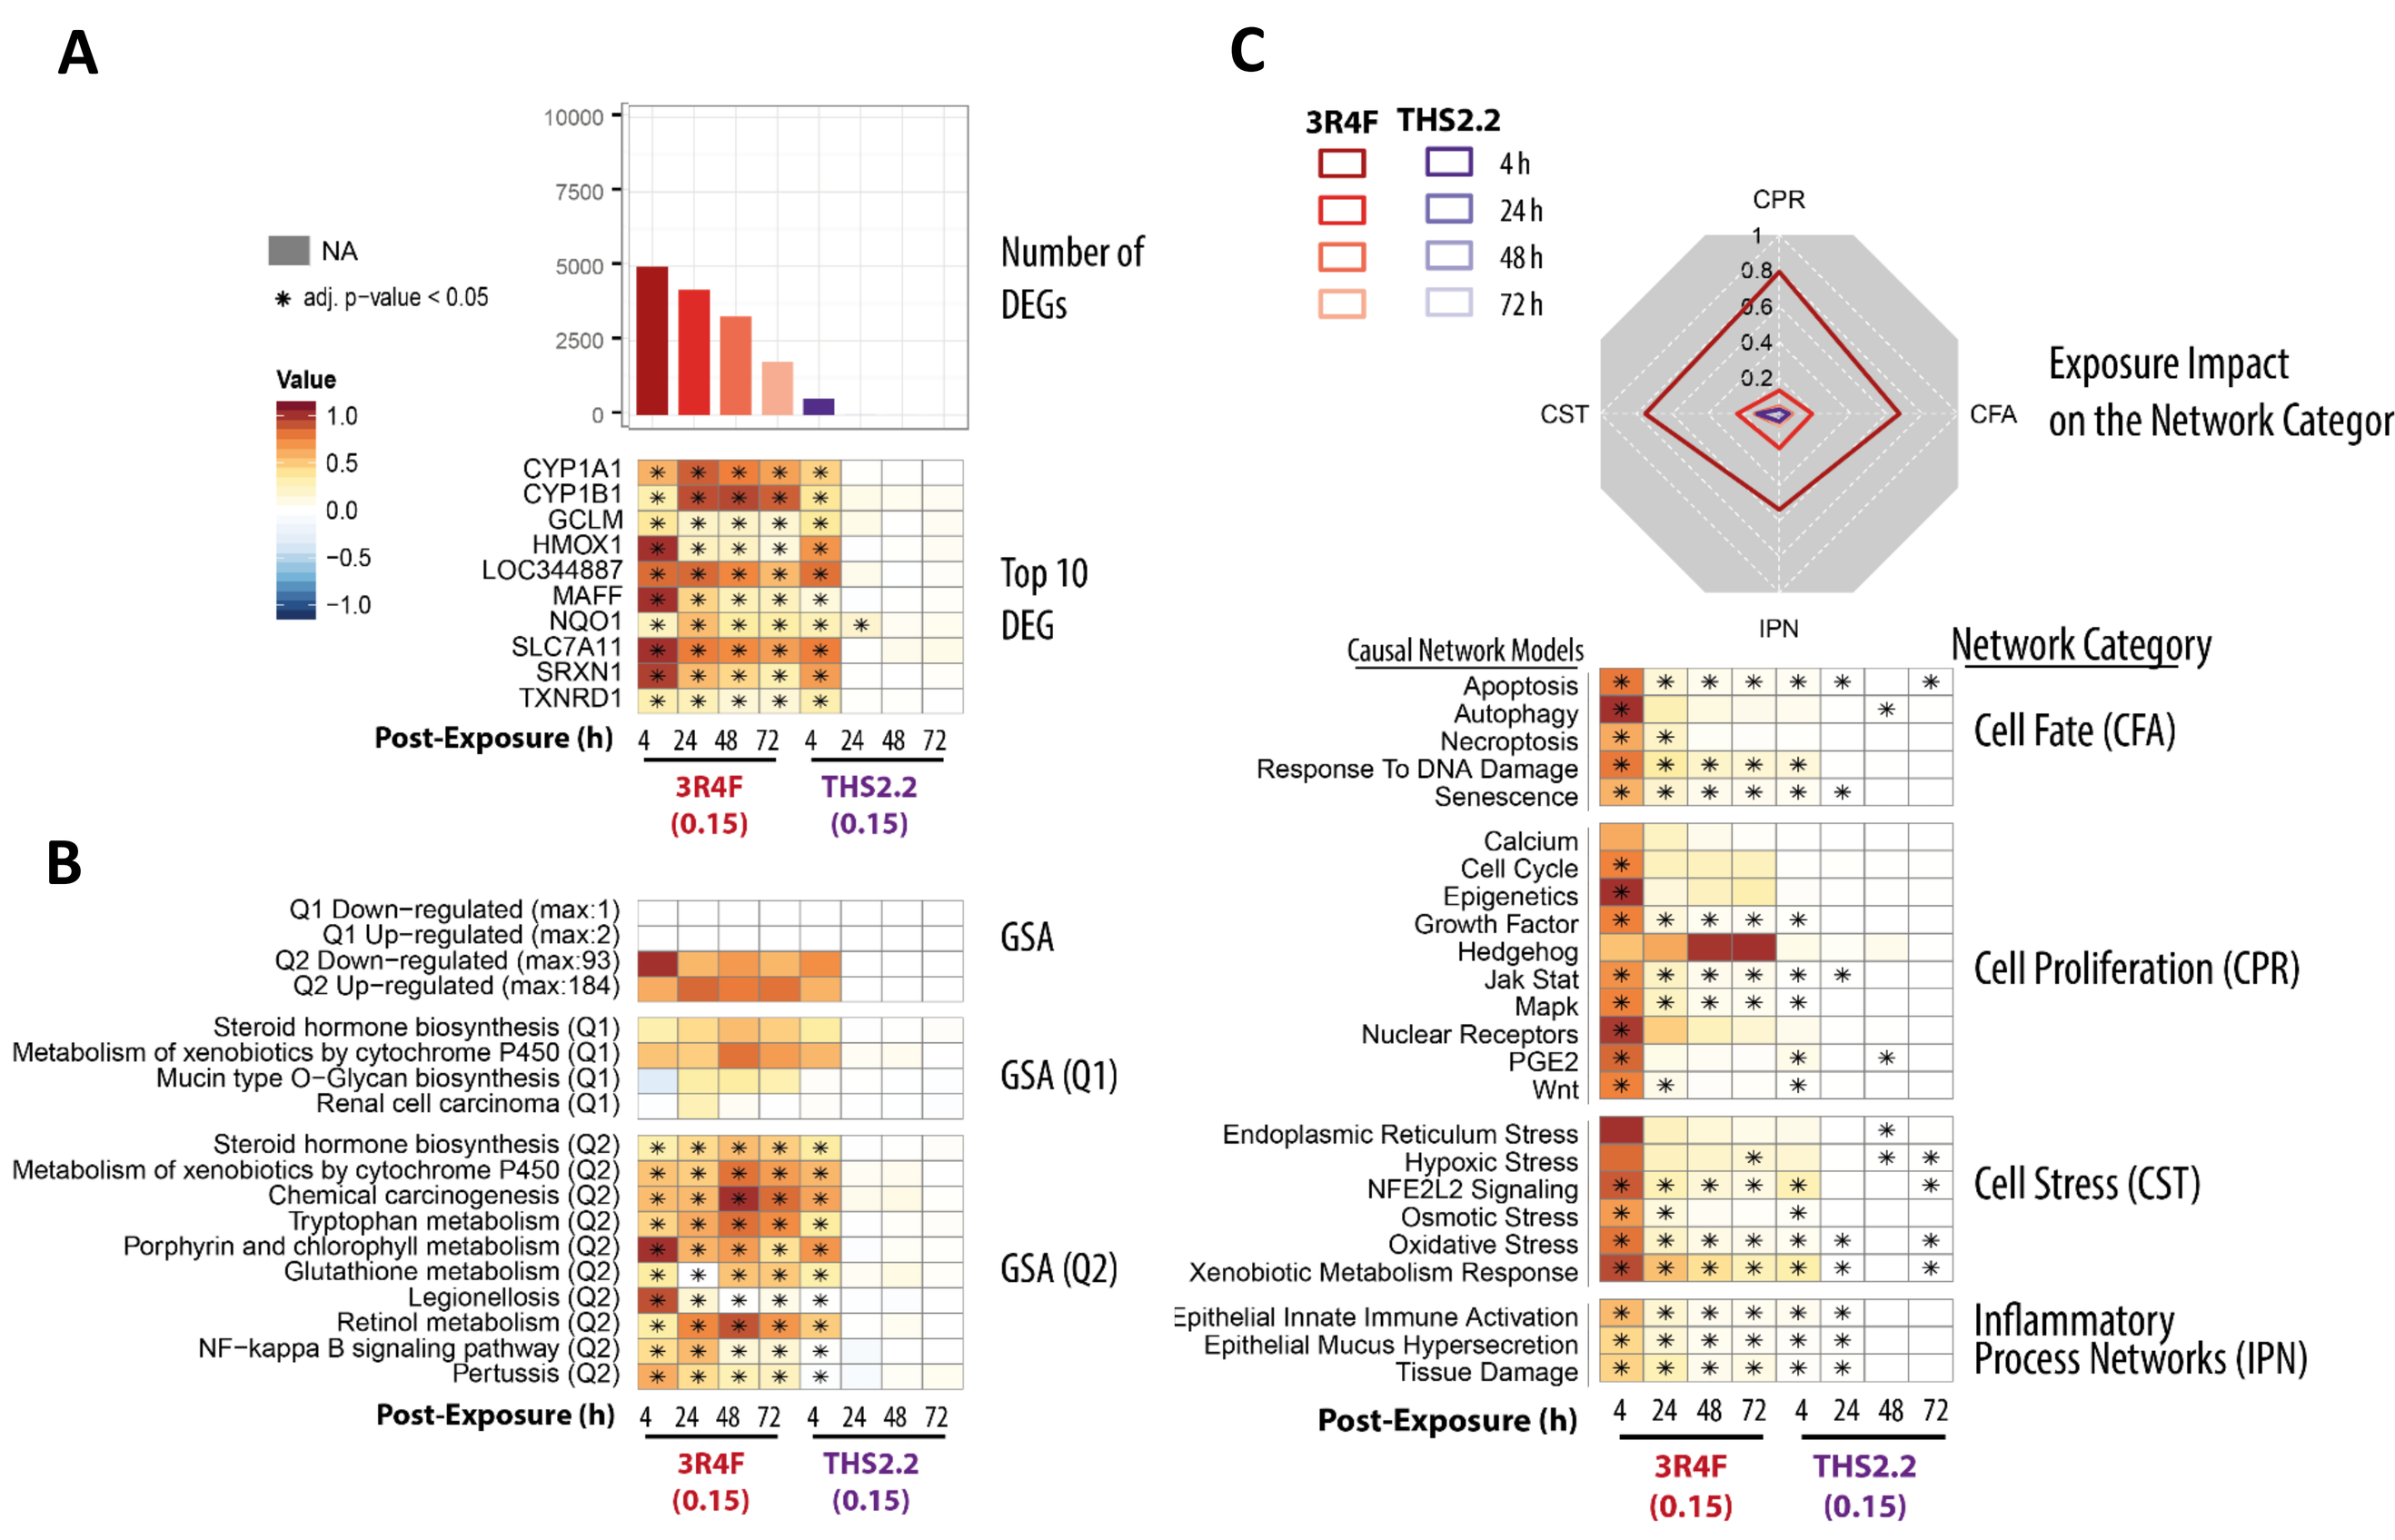
A
Number of DEGs
Top 10 DEG
CYP1A1
CYP1B1
GCLM
HMOX1
LOC344887
MAFF
NQO1
SLC7A11
SRXN1
TXNRD1
NA
adj. p-value < 0.05
Value
Post-Exposure (h) 4 24 48 72 4 24 48 72
3R4F (0.15)
THS2.2 (0.15)
B
Q1 Down−regulated (max:1)
Q1 Up−regulated (max:2)
Q2 Down−regulated (max:93)
Q2 Up−regulated (max:184)
GSA
Steroid hormone biosynthesis (Q1)
Metabolism of xenobiotics by cytochrome P450 (Q1)
Mucin type O−Glycan biosynthesis (Q1)
Renal cell carcinoma (Q1)
GSA (Q1)
Steroid hormone biosynthesis (Q2)
Metabolism of xenobiotics by cytochrome P450 (Q2)
Chemical carcinogenesis (Q2)
Tryptophan metabolism (Q2)
Porphyrin and chlorophyll metabolism (Q2)
Glutathione metabolism (Q2)
Legionellosis (Q2)
Retinol metabolism (Q2)
NF−kappa B signaling pathway (Q2)
Pertussis (Q2)
GSA (Q2)
C
3R4F THS2.2
4 h
24 h
48 h
72 h
CPR
CFA
IPN
CST
Exposure Impact on the Network Category
Causal Network Models
Network Category
Apoptosis
Autophagy
Necroptosis
Response To DNA Damage
Senescence
Cell Fate (CFA)
Calcium
Cell Cycle
Epigenetics
Growth Factor
Hedgehog
Jak Stat
Mapk
Nuclear Receptors
PGE2
Wnt
Cell Proliferation (CPR)
Endoplasmic Reticulum Stress
Hypoxic Stress
NFE2L2 Signaling
Osmotic Stress
Oxidative Stress
Xenobiotic Metabolism Response
Cell Stress (CST)
Epithelial Innate Immune Activation
Epithelial Mucus Hypersecretion
Tissue Damage
Inflammatory Process Networks (IPN)

Figure 4: Mechanistic investigation of the exposure impact based on the transcriptomic data
(A) Barplots showing the number of significantly differentially expressed genes (DEGs) across the exposure conditions (FDR-adjusted p-value < 0.05). The heatmaps indicate the expression profiles of the top ten genes (sorted first by the number of significant conditions and then by the mean of the absolute fold-changes). The log2(fold-changes) compared with the respective air control groups are color-coded and the statistical significance level is indicated (FDR-adjusted p-value). (B) Gene set analysis (GSA) was performed with the KEGG gene-set collection using absolute log2(fold-changes) as the gene-level and the mean as the gene-set level statistics. Significance with respect to the treatment effect (Q2, compared with the air control) and dominant effects of individual gene sets (Q1) was assessed with Benjamini-Hochberg based FDR adjustment (FDR adj. p-value < 0.05). The numbers of significantly up- and down regulated gene sets for Q1 and Q2 are shown in the top panel, and the top gene sets, first sorted by the number of significant conditions and then by their average absolute scores, are shown in the bottom panels. (C) The causal network enrichment approach for the analysis of the transcriptomic datasets. For each network category, the relative biological impact factor is shown in radar plots (CFA, Cell Fate; CPR, Cell Proliferation; CST, Cell Stress; IPN, Inflammatory Process Networks). These causal networks describe relevant physiologic cell responses[5] and have undergone a crowd-sourced review[6]. The heatmaps show the network perturbation amplitudes for each network in the collection, across all conditions. Figure adapted from Iskandar et al. (2017)[1].

Transcriptomics - Cellular Stress Response

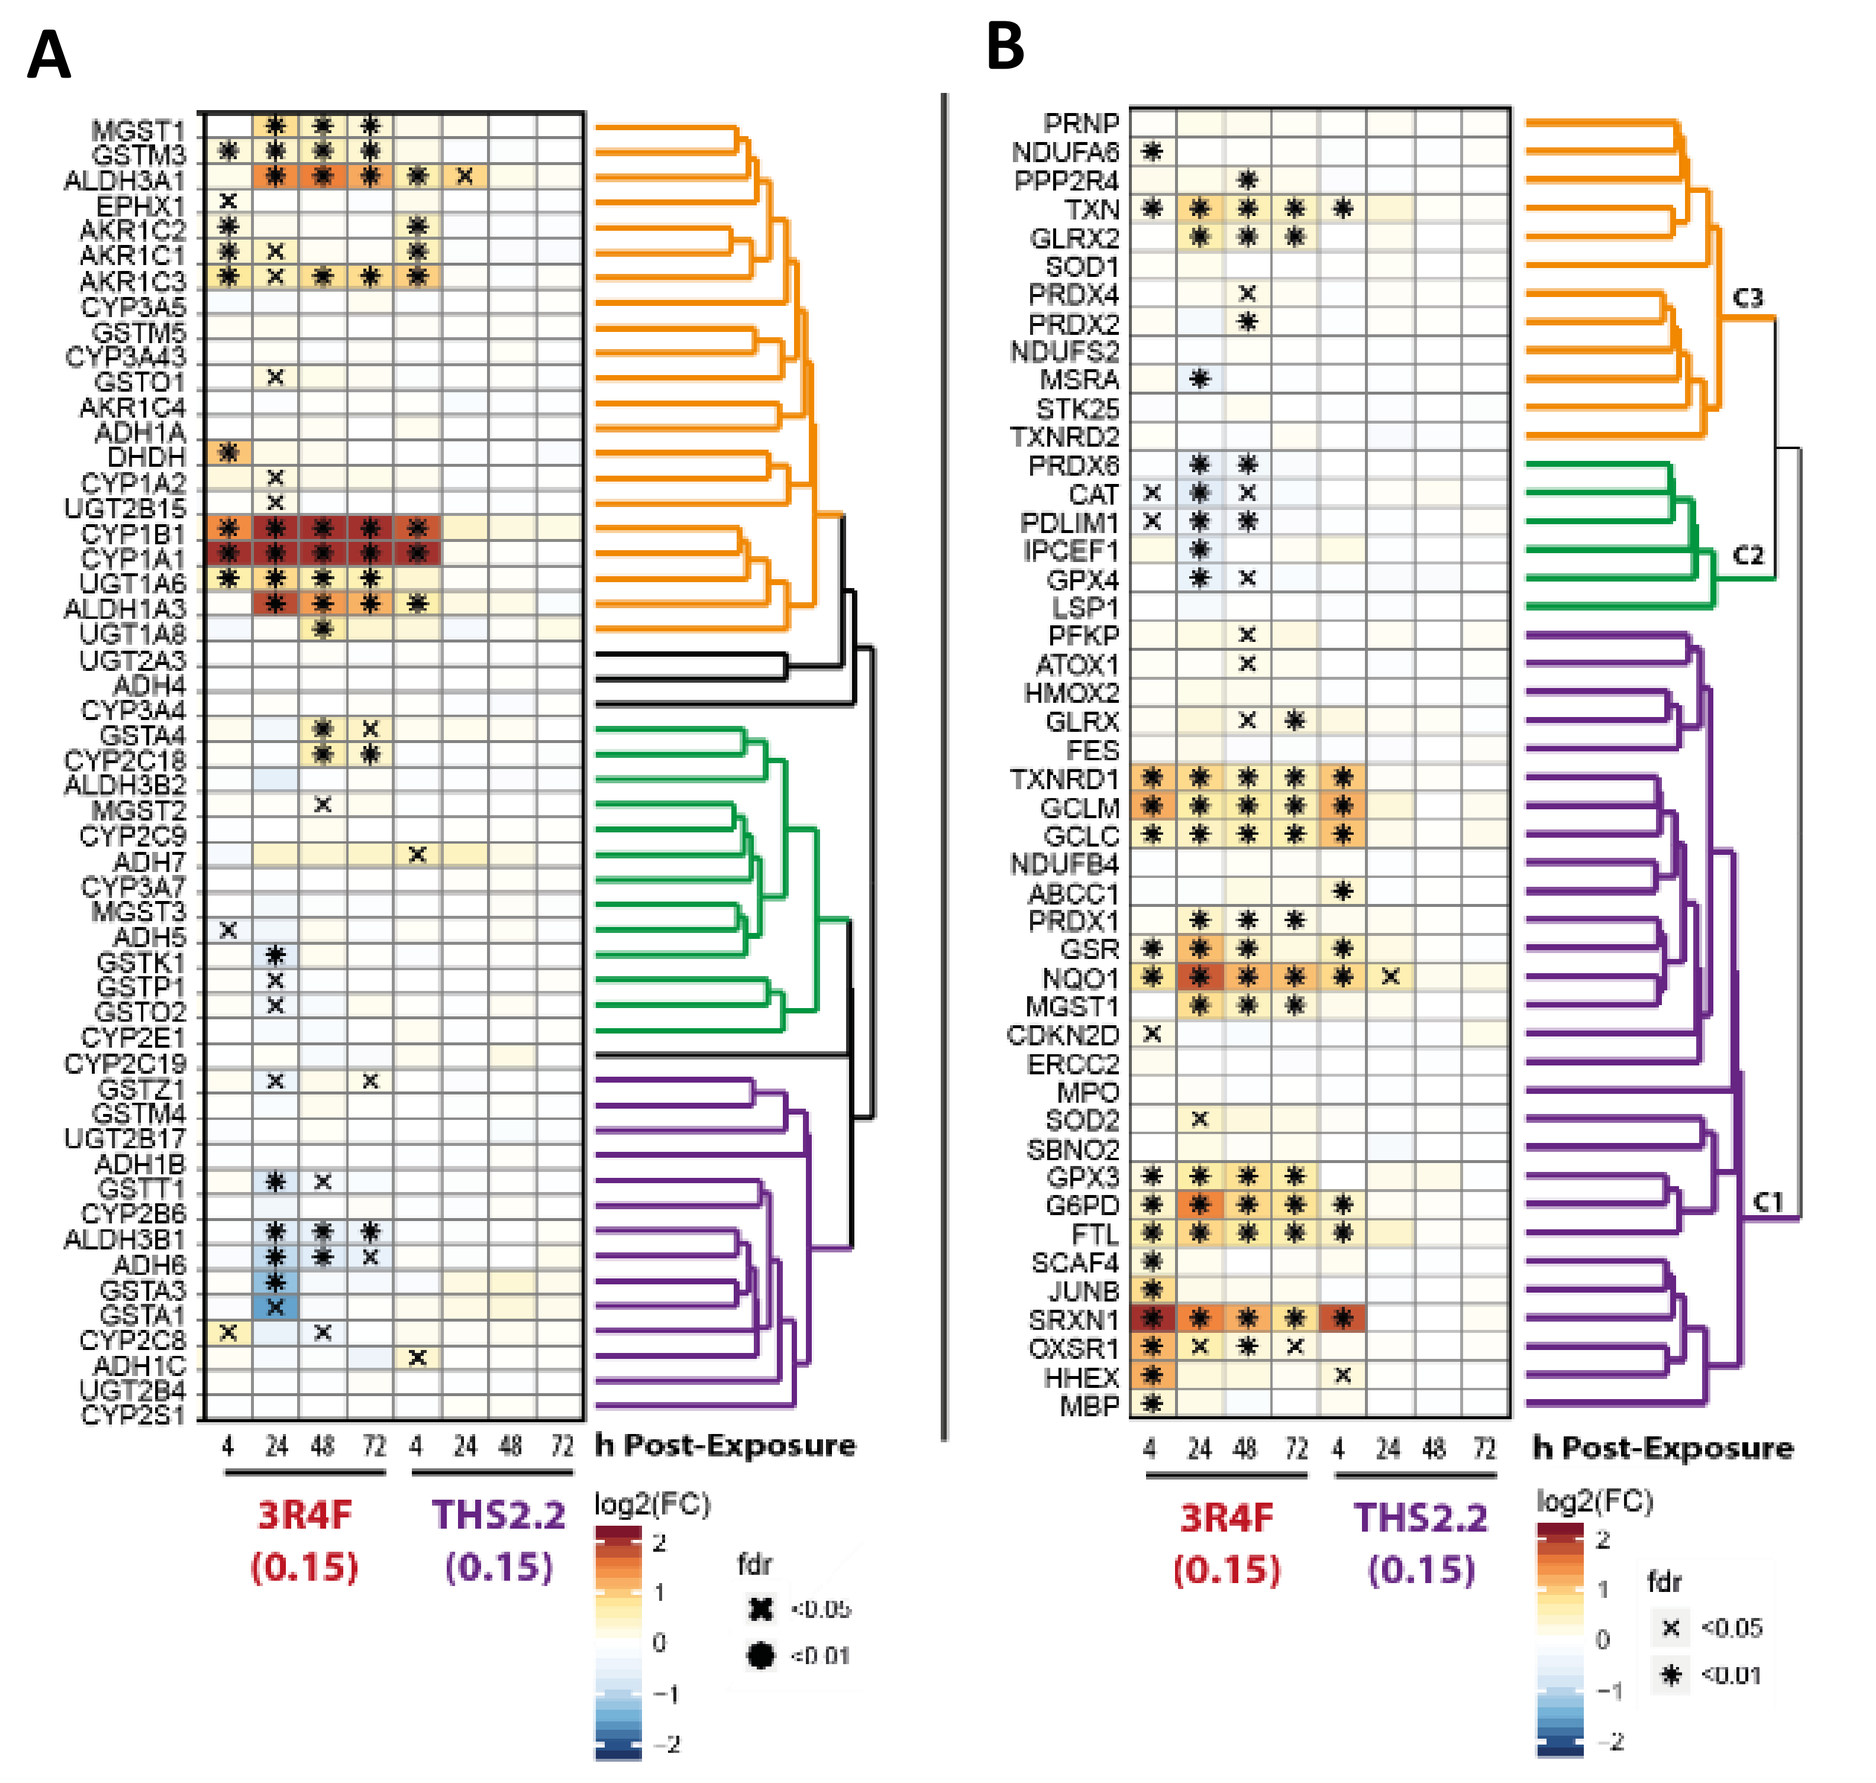
A
B
h Post-Exposure
3R4F (0.15)
THS2.2 (0.15)
log2(FC)
fdr
<0.05
<0.01
C1
C2
C3

Figure 5: Induction of xenobiotic metabolism and oxidative stress responses in the nasal organotypic epithelium cultures.
(A) Clustered gene expression matrix for genes of a xenobiotic metabolism gene set (KEGG collection, Metabolism of Xenobiotics by Cytochrome P450). The log2 fold-changes compared with the air control are color-coded and FDR-adjusted significance is indicated for the FDR-adjusted p-value < 0.05 (x) and < 0.01 () levels. Gene clustering based on the pair-wise correlation between the fold-changes and the clustering results are shown as a dendrogram (clusters are marked in different colors). (B) As in B, but for the oxidative stress response gene set (Reactive Oxygen Species Pathway of the hallmark collection of mSigDB). Figure adapted from Iskandar et al. (2017)[1].*

Targeted Proteomics – Cellular Stress and Pro-Inflammatory Responses

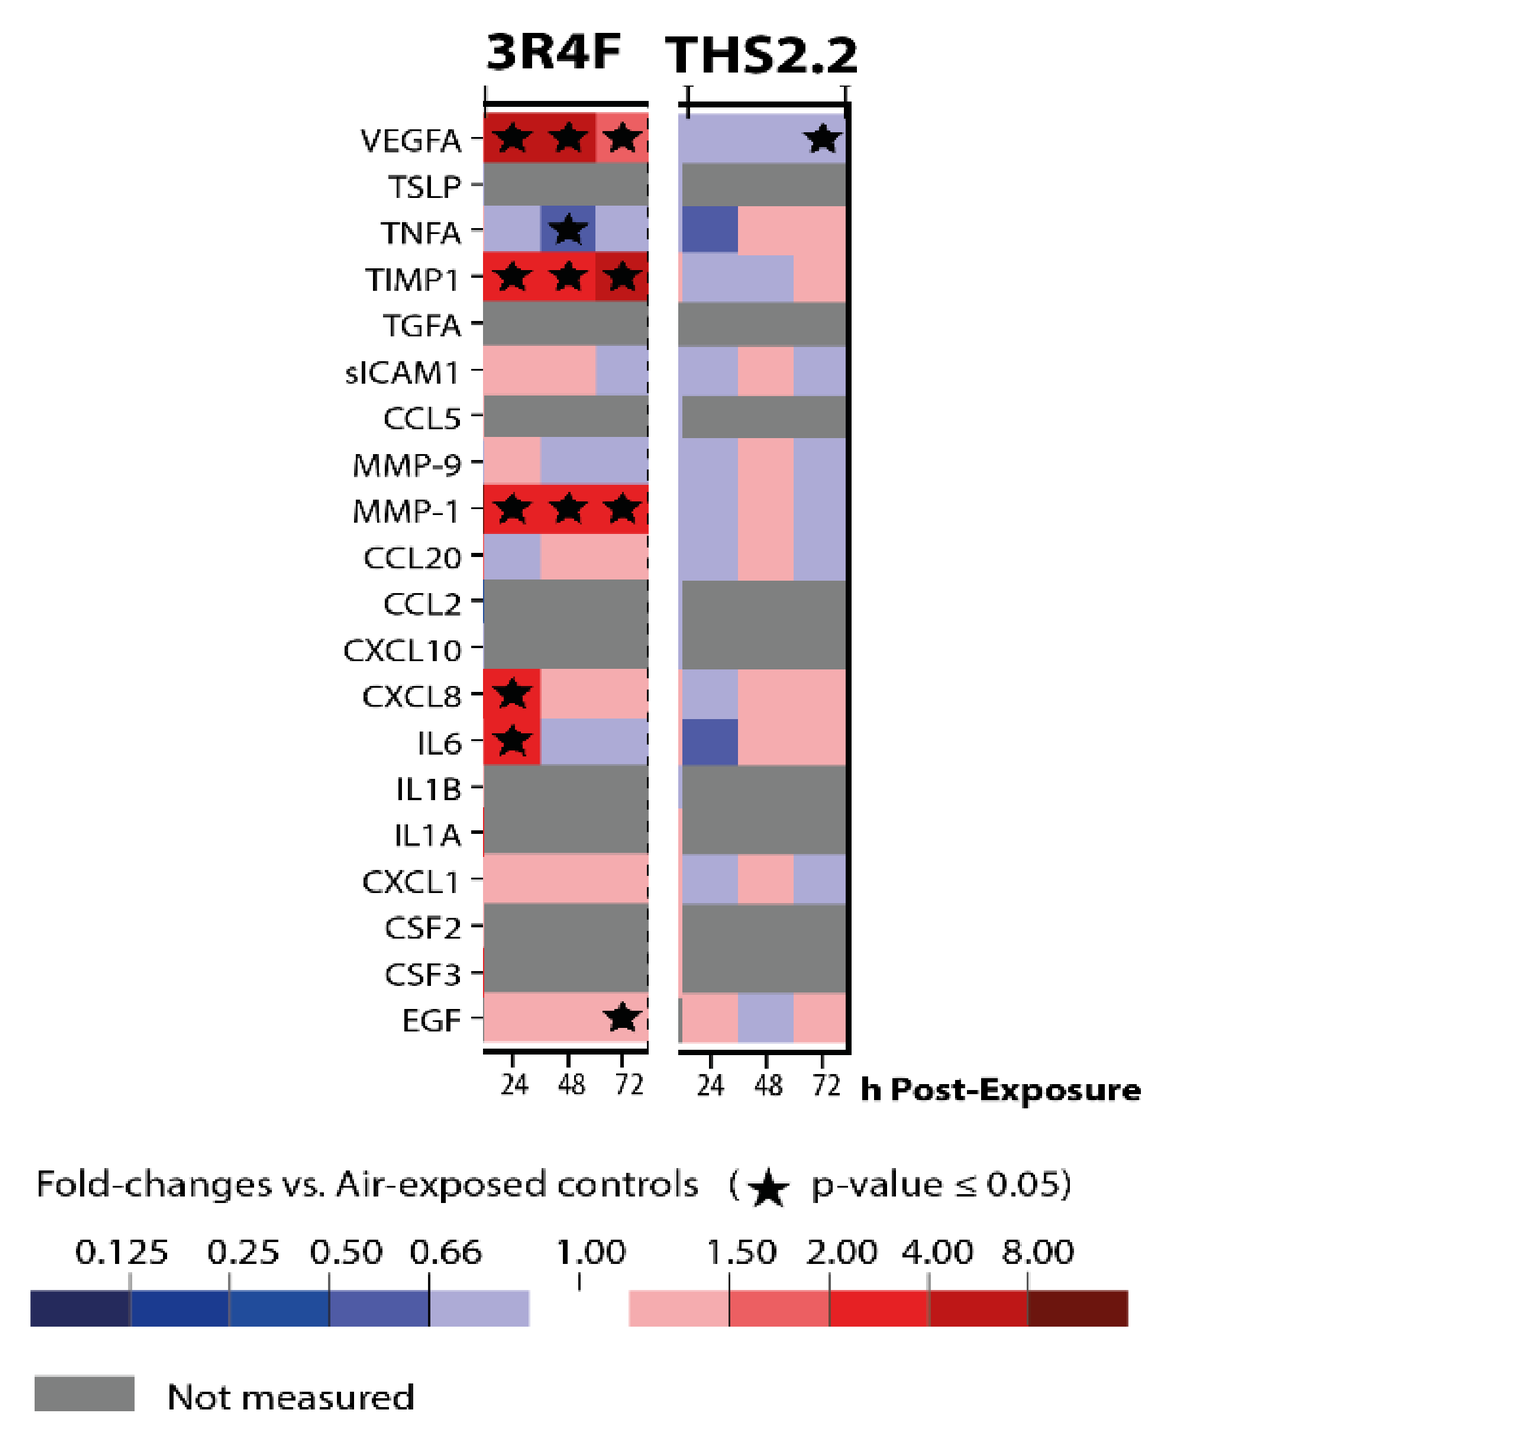
3R4F THS2.2
VEGFA
TSLP
TNFA
TIMP1
TGFA
sICAM1
CCL5
MMP-9
MMP-1
CCL20
CCL2
CXCL10
CXCL8
IL6
IL1B
IL1A
CXCL1
CSF2
CSF3
EGF
24 48 72 24 48 72 h Post-Exposure
Fold-changes vs. Air-exposed controls (★ p-value ≤ 0.05)
0.125 0.25 0.50 0.66 1.00 1.50 2.00 4.00 8.00
Not measured

Figure 6. Exposure-induced pro-inflammatory responses.
Multianalyte profiling (MAP) data for secreted pro-inflammatory mediators measured at various post-exposure time points. Figure adapted from Iskandar et al. (2017)[1].

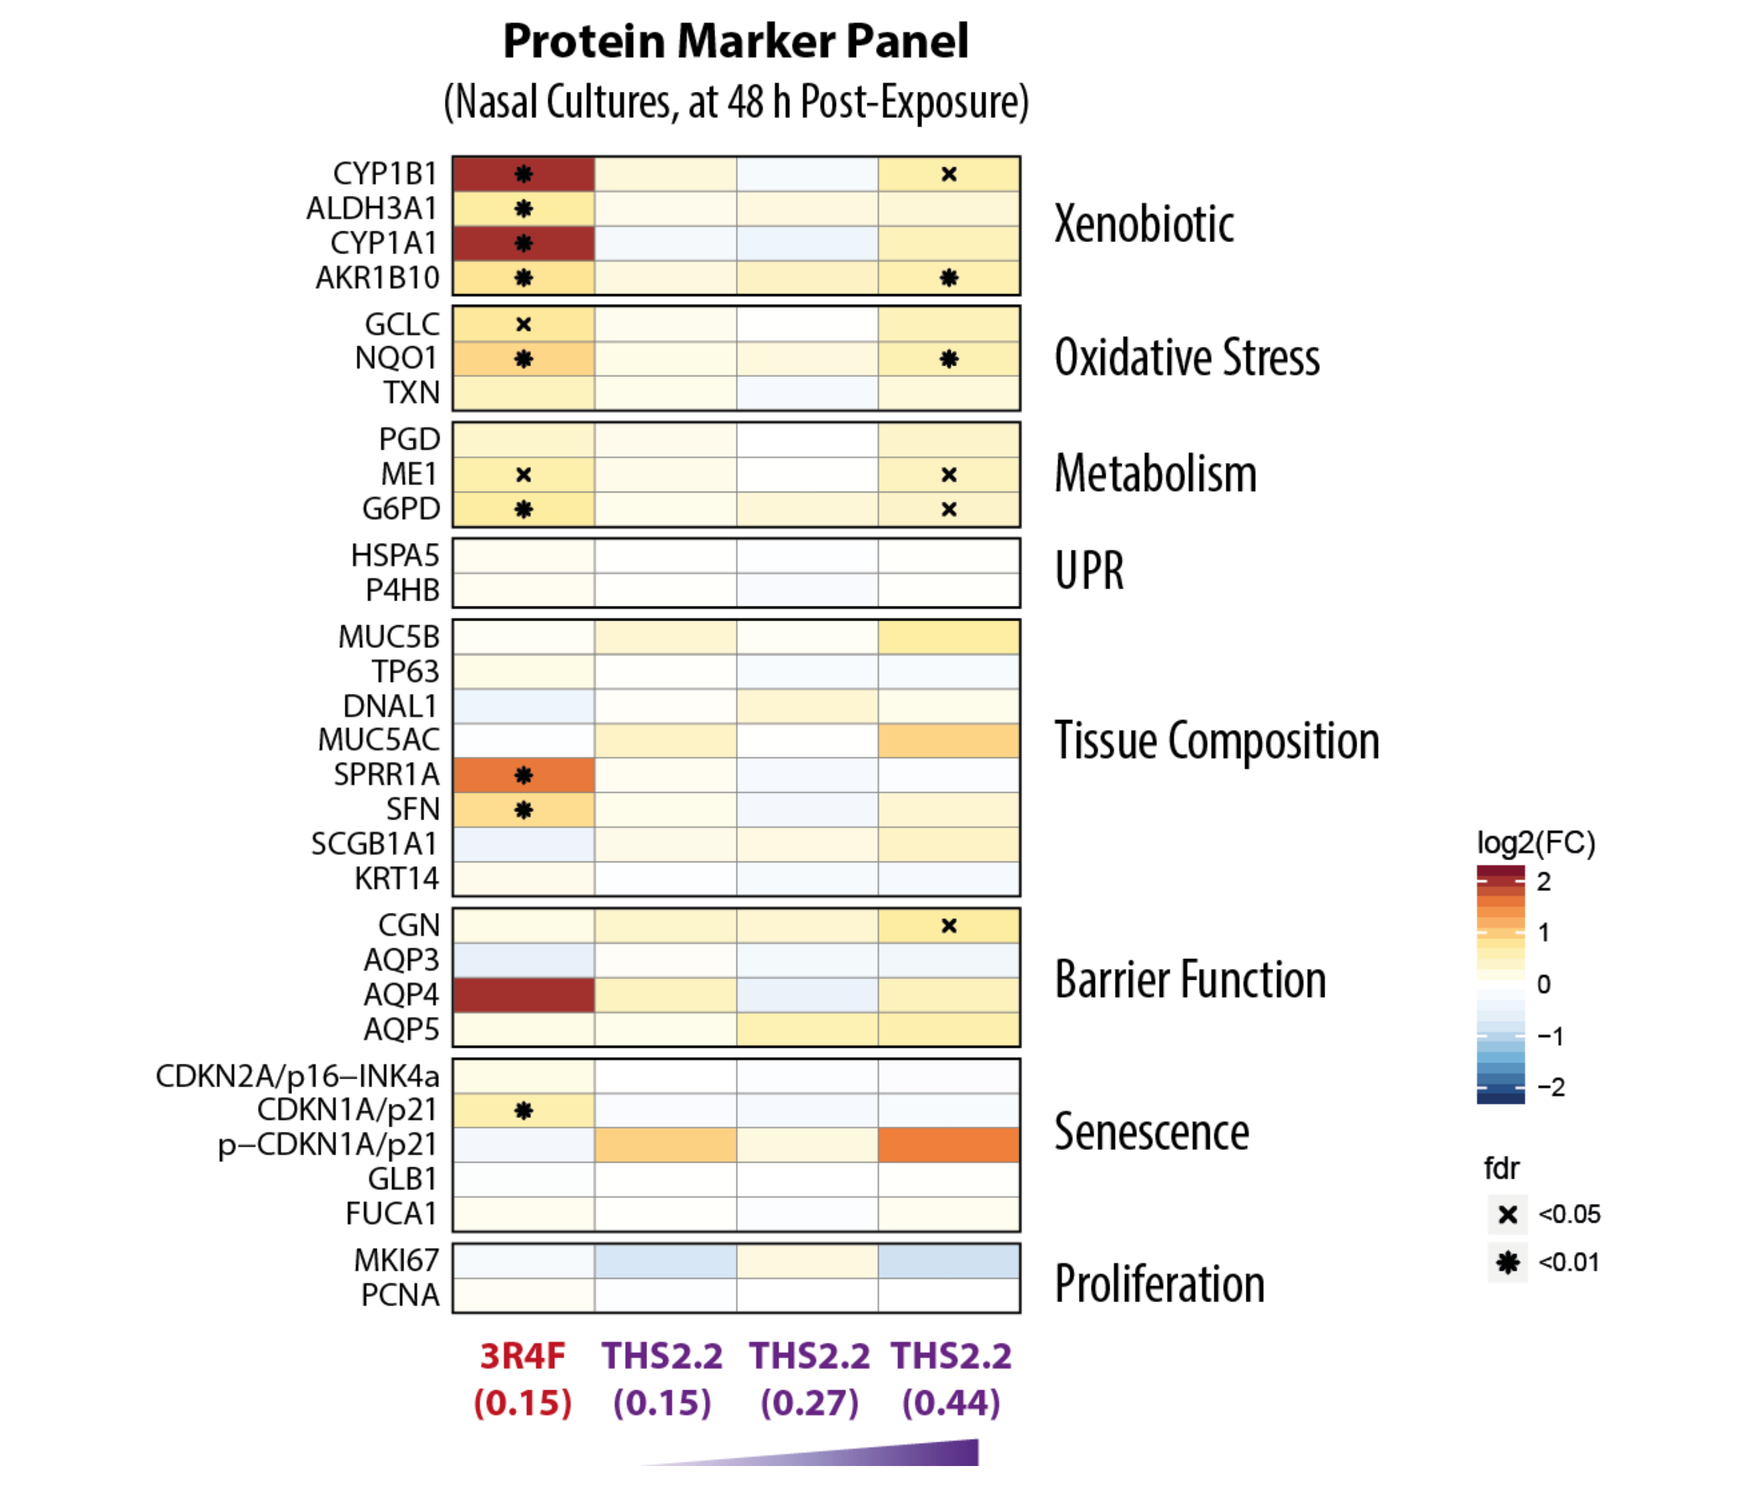
Protein Marker Panel
(Nasal Cultures, at 48 h Post-Exposure)
CYP1B1
ALDH3A1
CYP1A1
AKR1B10
Xenobiotic
GCLC
NQO1
TXN
Oxidative Stress
PGD
ME1
G6PD
Metabolism
HSPA5
P4HB
UPR
MUC5B
TP63
DNAL1
MUC5AC
SPRR1A
SFN
SCGB1A1
KRT14
Tissue Composition
CGN
AQP3
AQP4
AQP5
Barrier Function
CDKN2A/p16−INK4a
CDKN1A/p21
p−CDKN1A/p21
GLB1
FUCA1
Senescence
MKI67
PCNA
Proliferation
log2(FC)
fdr
<0.05
<0.01
3R4F (0.15)
THS2.2 (0.15)
THS2.2 (0.27)
THS2.2 (0.44)

Figure 7. Alterations of proteins in the nasal organotypic cultures following exposure measured by parallel-reaction monitoring (PRM).
The log2(fold-changes) compared with the air control are color-coded and the FDR-adjusted p-values are indicated (UPR, unfolded protein response). Figure from Iskandar et al. (2017)[1].

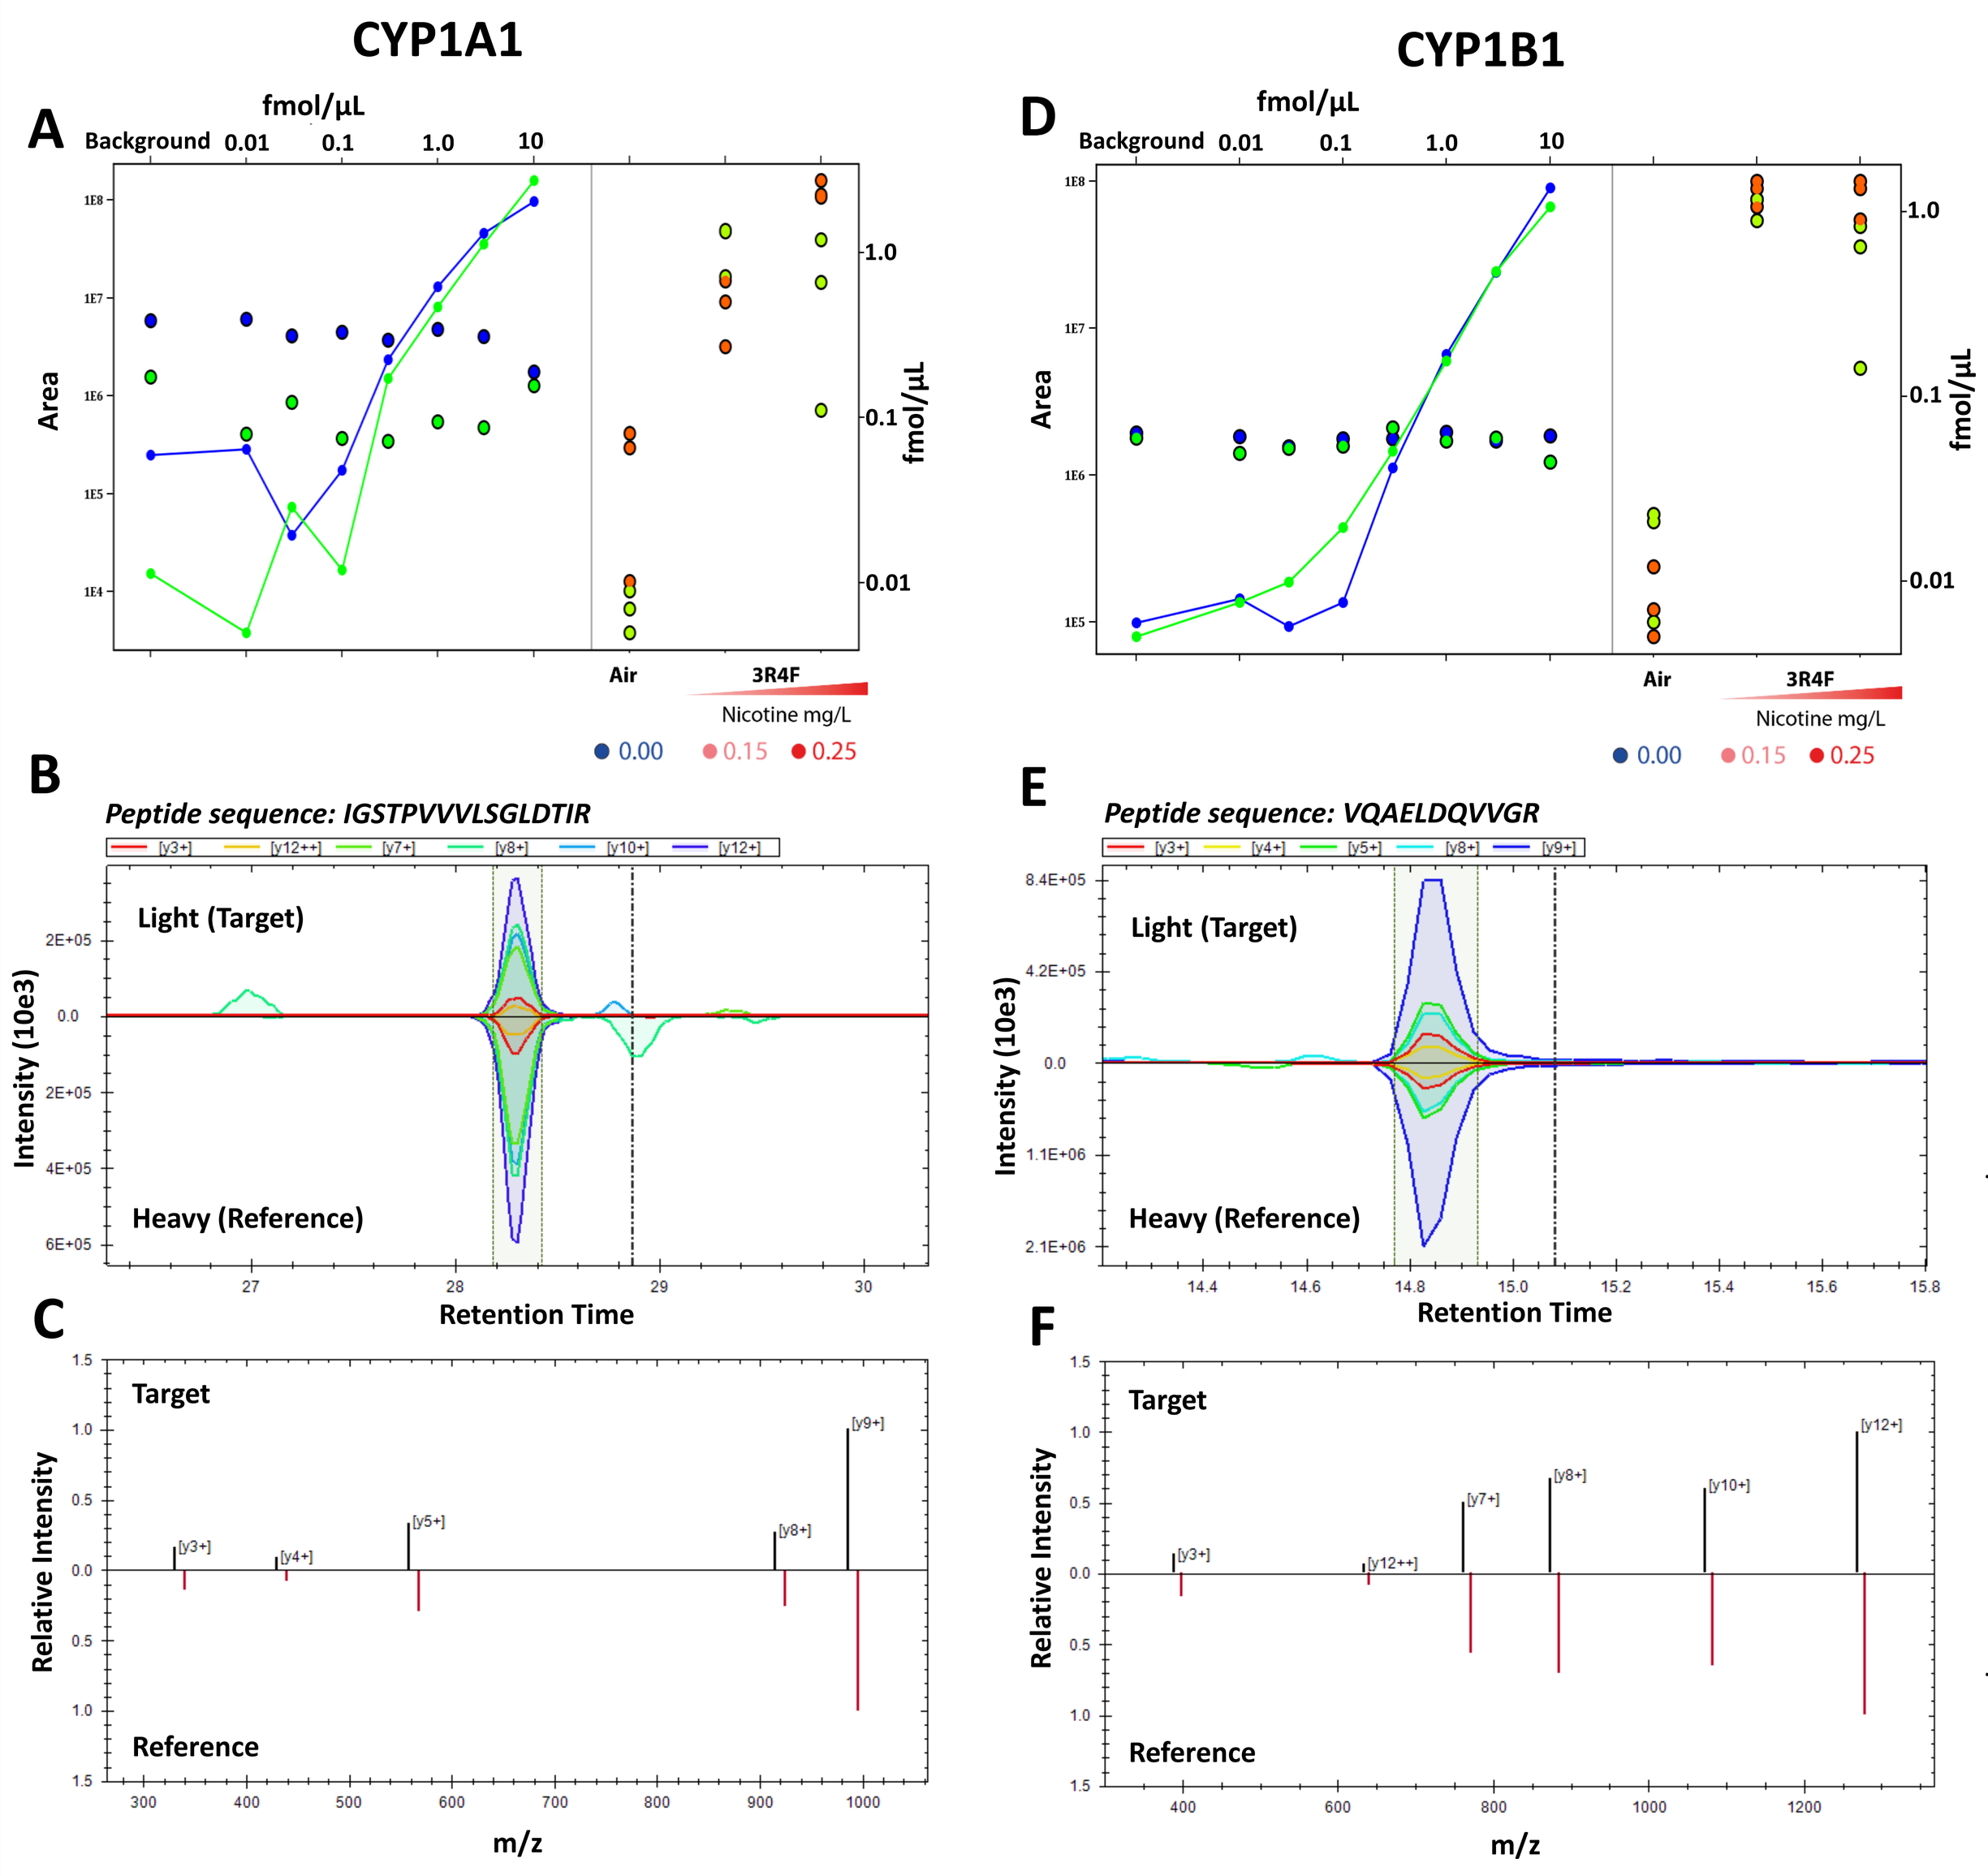
CYP1A1
CYP1B1
A
D
fmol/µL
Background 0.01 0.1 1.0 10
Area
Air
3R4F
Nicotine mg/L
0.00 0.15 0.25
Heavy (Reference)
Replicate 1
Replicate 2
Light (Target)
Replicate 1 Pool Study samples
Replicate 2 Pool Study samples
B
Peptide sequence: IGSTPVVVLSGLDTIR
E
Peptide sequence: VQAELDQVVGR
Light (Target)
Heavy (Reference)
Intensity (10e3)
Retention Time
C
F
Target
Reference
Relative Intensity
m/z

Figure 8. Representative graphs for expression level determination of the two cytochrome P450 proteins CYP1A1 and CYP1B1.
(A) CYP1A1 stable isotope labelled (SIL) peptide dilution curve experiments in nasal culture background and determined target protein concentration in response to 3R4F CS treatment. (B) CYP1A1 Transition ion-traces of peptide IGSTPVVVLSGLDTIR used for protein quantification. (C) Intensity correlation chart of fragment ions from SIL peptide (Reference) and target peptide for CYP1A1 specific peptide shown in B. (D) CYP1B1 stable isotope labelled (SIL) peptide dilution curve experiments in nasal culture background and determined target protein concentration in response to 3R4F CS treatment. (E) CYP1B1 Transition ion-traces of peptide VQAELDQVVGR used for protein quantification. (F) Intensity correlation chart of fragment ions from SIL peptide (Reference) and target peptide for CYP1B1 specific peptide shown in E.

Conclusions

- Lower cytotoxicity was observed in the nasal organotypic cultures following exposure to THS2.2 aerosol compared with 3R4F CS.
- The causal network enrichment approach demonstrated a broad mechanistic impact of 3R4F CS in the nasal organotypic culture model including xenobiotic, oxidative stress, and inflammatory responses. At comparable nicotine concentrations, THS2.2 aerosol elicited reduced and more transient effects on these processes than CS.
- Demonstrated applicability of the Systems Toxicology approach to quantify and compare the effects of 3R4F CS and THS 2.2 aerosol exposure on the level of pertinent biological mechanisms, including cellular stress and pro-inflammatory responses.
- Developed a 31-plex protein targeted panel for PRM relevant to biological processes perturbed by 3R4F CS exposure such as Xenobiotic and oxidative stress response, metabolism, unfolded protein response, barrier function, senescence and cell proliferation.
- Exemplified how targeted proteomics using PRM and antibody-based multi-analyte-profiling can strengthen the conclusions from other endpoints: The targeted marker panels further confirmed the reduced cellular stress responses elicited by THS2.2 aerosol compared with 3R4F CS in the nasal organotypic culture model.

References
[1] Iskandar et al. (2017), Systems Toxicology Meta-Analysis of In Vitro Assessment Studies: Biological Impact of a Candidate Modified-Risk Tobacco Product Aerosol Compared with Cigarette Smoke on Human Organotypic Cultures of the Aerodigestive Tract, *Toxicol. Res.*, Accepted, doi: 10.1039/C7TX00047B
[2] Iskandar et al. (2016), 3-D nasal cultures: Systems toxicological assessment of a candidate modified-risk tobacco product, *ALTEX*, DOI: 10.14573/altex.1605041.
[3] Family Smoking Prevention and Tobacco Control Act (2009). Public Law No. 111-131 (June 22, 2009).
[4] Escher et al. (2012), Using iRT, a normalized retention time for more targeted measurement of peptides. Proteomics. 2012; 12(8):1111-1121.
[5] Boué et al. (2015), Causal biological network database: a comprehensive platform of causal biological network models focused on the pulmonary and vascular systems, Database, bav030
[6] The sbv IMPROVER project (2015), Enhancement of COPD biological networks using a web-based collaboration interface, *F1000Research*, 4:32.

PMI SCIENCE
PHILIP MORRIS INTERNATIONAL

HUPO 2017, Dublin

September 17-20, 2017

Competing Financial Interest

All authors are employees of Philip Morris International (PMI) or worked for PMI under contractual agreements. PMI is the sole source of funding and sponsor of this project.