Delivery of glycerol-based aerosols in the Vitrocell®24/48 aerosol exposure system

Sandro Steiner, Shoaib Majeed, Gilles Kratzer, Mounir Rhouma, Quentin Dutertre, Grégory Vuillaume, Arno Knorr, Julia Hoeng, Stefan Frentzel
sandro.steiner@pmi.com

Philip Morris International R&D, Philip Morris Products S.A.
(part of Philip Morris International group of companies)

Introduction and Objectives

The increasing market presence of electronic cigarettes (e-cigarettes) requires establishing adequate *in vitro* methodologies for their appropriate toxicological assessment. In this context, the possibility of conducting controlled *in vitro* aerosol exposures resulting in reproducible dose delivery becomes very important. Moreover the delivered doses need to be relevant for *in vivo* exposures and comparable to *in vitro* reference exposures - commonly exposures to cigarette smoke.

We performed an experimental system characterization of the Vitrocell®24/48 aerosol exposure system for its use in conjunction with aerosols generated from e-cigarettes in order to assess the suitability of the system for this application *(1)*. A glycerol model aerosol with a particle size distribution representative of aerosols generated by e-cigarettes was used for exposing small volumes of phosphate-buffered saline placed into the exposure chambers of the Vitrocell®24/48 as surrogates for cell cultures. Disodium fluorescein added as a tracer in the aerosol allowed the exact particle mass delivery to each exposure chamber to be quantified fluorometrically with the ultimate goal of determining the dose delivery as function of aerosol dilution, the uniformity of delivery to replica positions and the repeatability of exposures. The observed dose delivery was in addition put into relation to the delivery of smoke generated from 3R4F research cigarettes.

Methods

Aerosol generation:

Fluorescent glycerol aerosols were generated in a TSI Condensation Monodisperse Aerosol Generator (TSI 3475 CMAG). An aerosol of disodium fluorescein (DSF) nuclei is bubbled through heated glycerol (160°C) and cooling of the resulting vapor-nuclei mixture results in the condensation of glycerol on the DSF. For the present work, an aerosol with a mean particle size of 830 nm and a geometric standard deviation of 1.29 was generated (Figure 1).

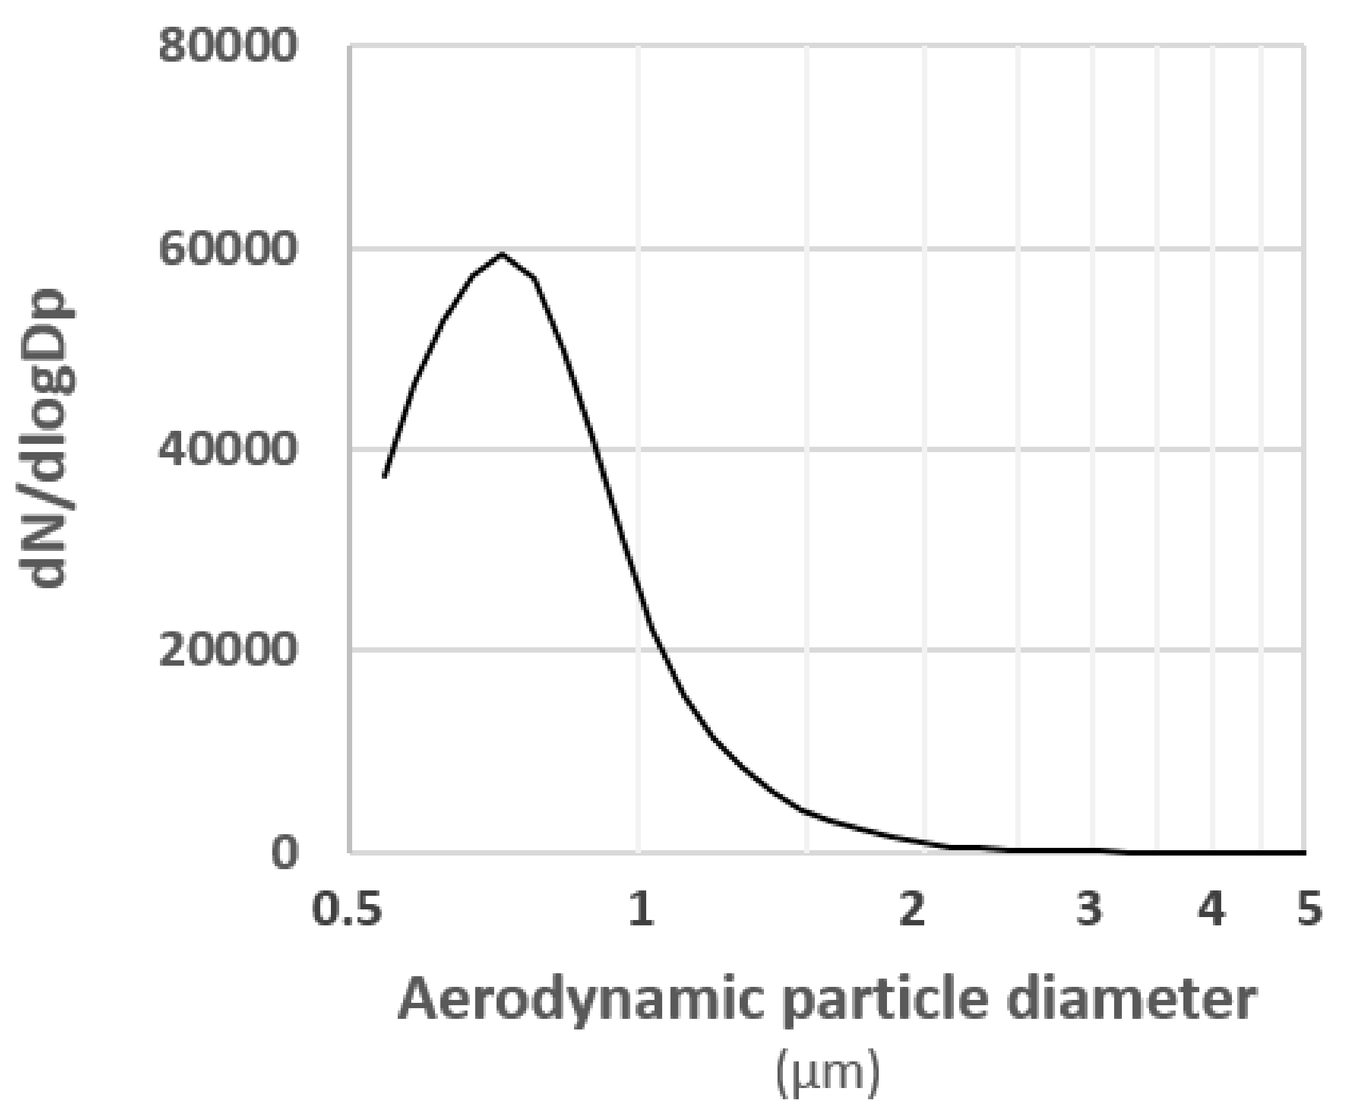
dN/dlogDp
Aerodynamic particle diameter
(µm)

Figure 1: *Particle size distribution of the glycerol model aerosol.*

Aerosol exposure:

Phosphate buffered saline samples (PBS, 100 µL samples) present in cell culture inserts (Greiner Bio-One) in the exposure chambers of the Vitrocell®24/48 aerosol exposure system (Vitrocell GmbH, Waldkirch, Germany) were exposed to glycerol aerosol. Exposures were conducted under the following basic exposure conditions (the working principle of the Vitrocell system and the applied dilution air flows are shown in Figure 2):

- An aerosol flow rate of 413 mL/min was supplied to the Vitrocell system, corresponding to a puff of 55 mL provided during 8 seconds
- The total exposure duration was 28 minutes
- The aerosol was diluted serially by adding a cumulative dilution air flow rate of 100, 200, 500, 1000, 1500, 2000 and 3000 mL/min at dilution row 1 to 7 (resulting in 81, 67, 45, 29, 22, 17 and 12% aerosol, see Figure 2)
- The flow rate through exposure trumpets was set to 2mL/min for all positions

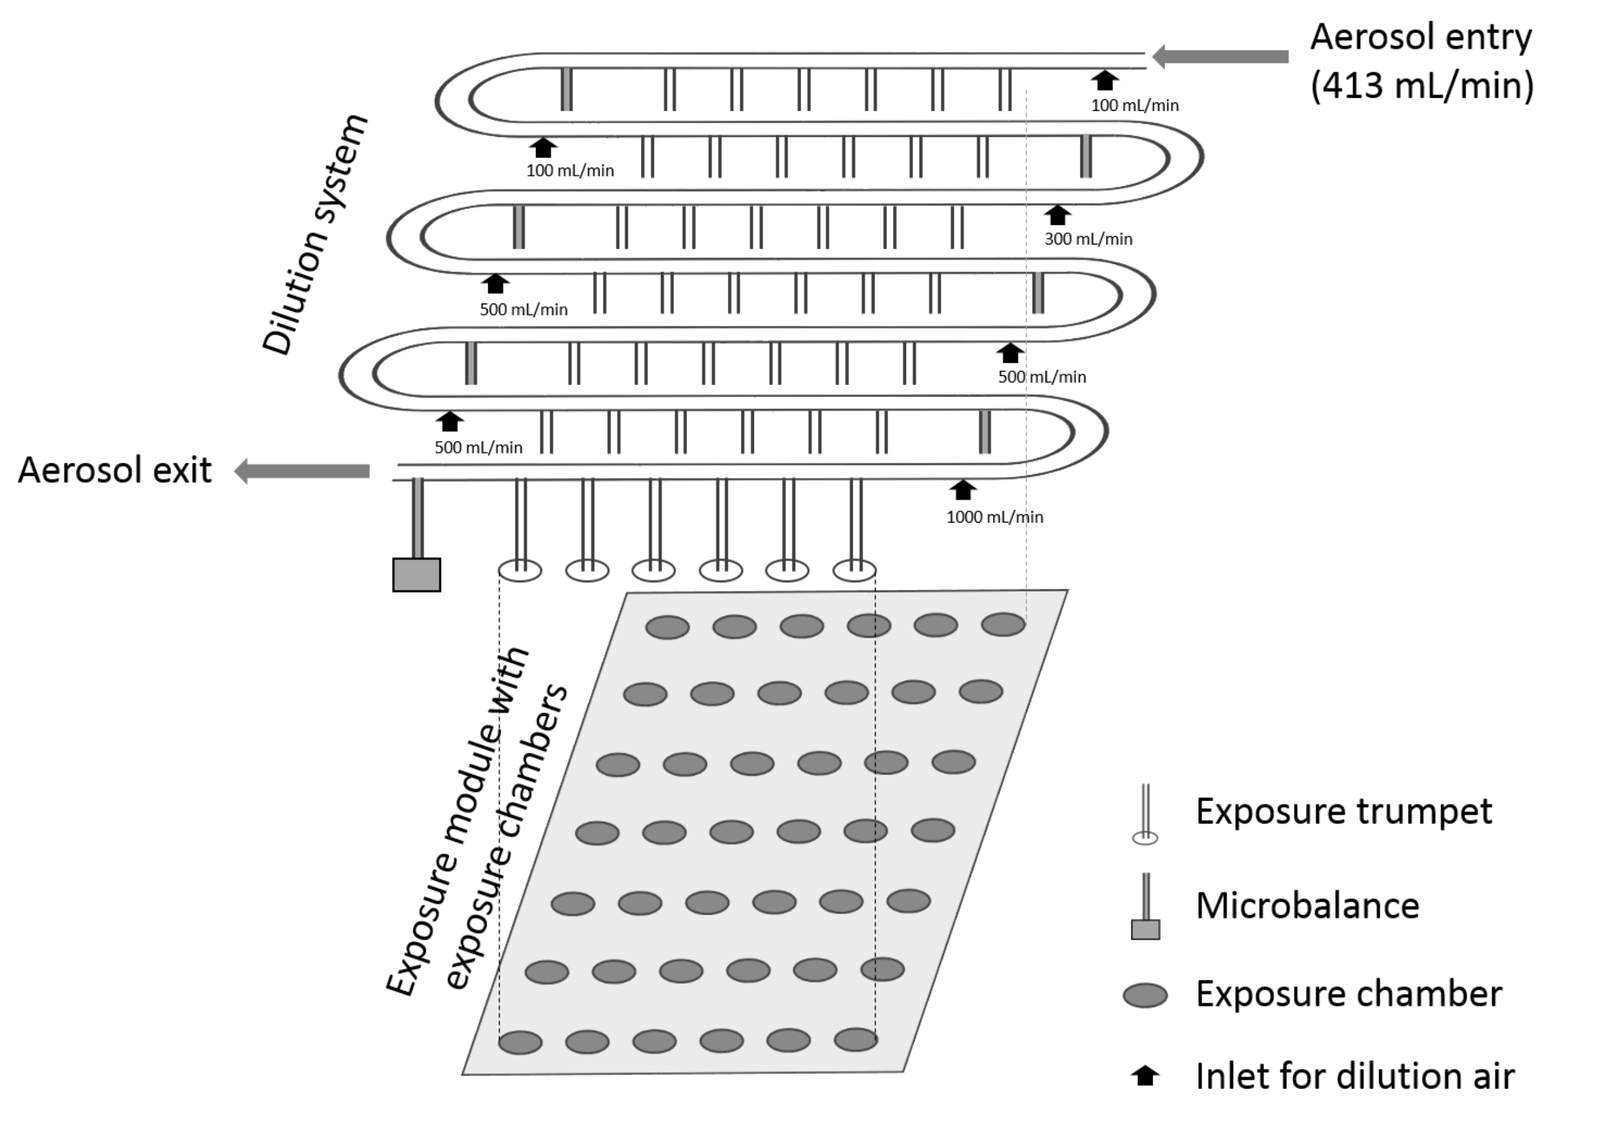
Aerosol entry
(413 mL/min)
Dilution system
Aerosol exit
Exposure module with exposure chambers
Exposure trumpet
Microbalance
Exposure chamber
Inlet for dilution air

Figure 2: *Working principle of the Vitrocell®24/48 aerosol exposure system. the sites and flow rates of dilution air application are indicated.*

Aerosol characterization:

Before, in the middle of (after 14 minutes) and after each exposure, the aerosol flow through the Vitrocell® 24/48 aerosol exposure system was interrupted and aerosol characterization was performed:

- The aerosol was trapped on Cambridge filter pads (CFP, Borgwaldt KC, Hamburg, Germany). The trapped aerosol was weighed and eluted from the filters in PBS
- The particle size distribution was measured using a TSI Aerodynamic Particle Sizer (TSI 3321 APS)

The Physical setup of the instrumentation used for aerosol characterization and test exposures is shown in Figure 3.

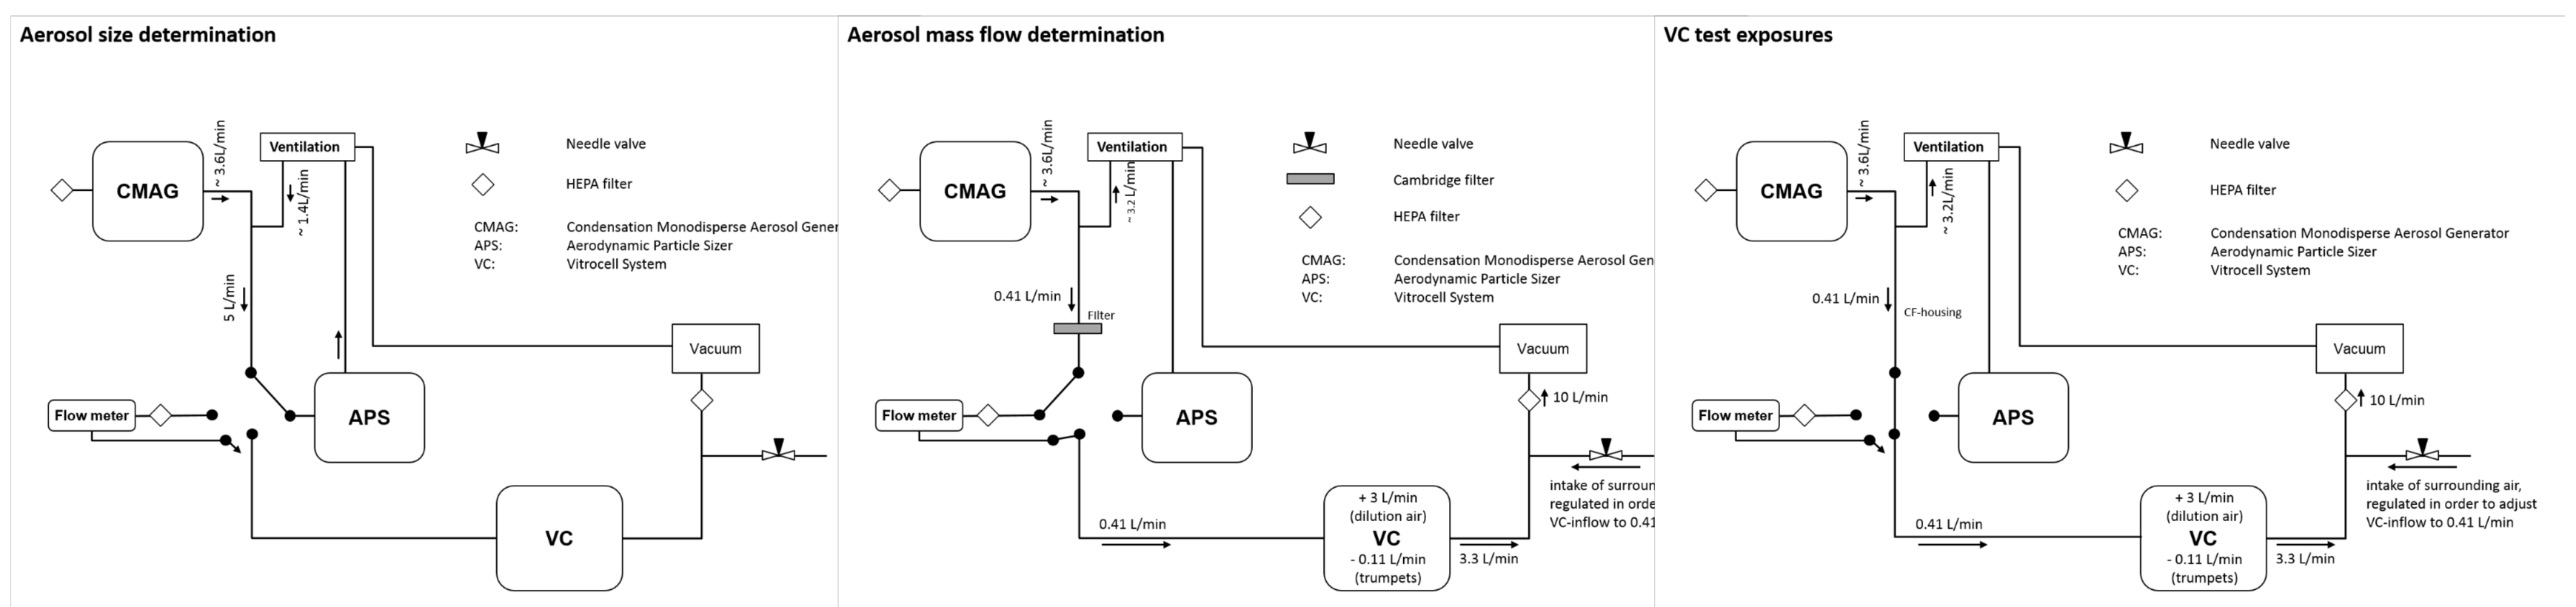
Aerosol size determination
Aerosol mass flow determination
VC test exposures
CMAG
Ventilation
APS
Vacuum
Flow meter
VC
Needle valve
Cambridge filter
HEPA filter

Figure 3: *Physical setup of the instrumentation used for aerosol characterization and test exposures*

Sample processing and analysis

For characterizing the exposures, the DSF fluorescence present in the PBS samples retrieved from cell culture inserts and from CFPs was quantified in a Fluostar Omega Microplate reader (BMG Labtech, Ortenberg, Germany) at an excitation wavelength of 485 nm and an emission wavelength of 520 nm.

The total particle mass delivery to individual PBS samples and the particle delivery efficiency were calculated according to equations 1 and 2. The particle delivery efficiency was thereby corrected for dilution in order to allow a direct comparison between individual dilution rows.

$$Total\ Particle\ mass\ delivery\ (position\ X) = Disodium fluorescein\ mass\ (position\ X)\ x\ \frac{Particle\ mass}{Disodium\ fluorescein\ mass} \qquad \text{Eq. 1}$$

$$Particle\ delivery\ efficiency = \frac{Particle\ mass\ delivery\ to\ position\ X}{Total\ particle\ mass\ flow\ passing\ the\ system}\ x\ \frac{1}{applied\ aerosol\ concentration} \qquad \text{Eq. 2}$$

Comparison to exposures using cigarette smoke

3R4F reference cigarettes (University of Kentucky) were smoked according to the Health Canada smoking regime (55 mL puffs taken every 30 seconds per cigarette *(2)*, puff release duration of eight seconds) and the generated smoke was used for exposing PBS samples in the Vitrocell®24/48. Settings and operation mode of the system were thereby identical to what is described for the glycerol aerosols. Nicotine is predominantly delivered to the respiratory tract via the particulate fraction of 3R4F smoke *(3)*, and was therefore chosen as reference smoke constituent for calculating particle delivery efficiencies (according to Eq. 2). Nicotine deposited in the PBS samples was quantified by high performance liquid chromatography coupled to tandem mass spectroscopy. Particle delivery efficiencies were calculated on the basis of 1.6 mg of total nicotine per cigarette (*(4),* smoke characterization was not performed)

Results

Dosing accuracy and repeatability of exposures

Figure 4 shows the particle mass delivery to individual dilution rows (averages over the six replica positions per row) as measured after each experimental repetition and as average over all repetitions. Although the correlation between dilution and delivery is not fully linear, the delivered particle doses reflect the applied aerosol concentration. The repeatability of exposures is high; except for at the highest and lowest aerosol concentrations all data points are located within a range of average ± 15%.

Uniformity of particle delivery

Figure 5 shows the particle mass delivery to each individual exposed cell culture insert after correction for dilution (division by the aerosol concentration). The uniformity of delivery to individual positions is not the same at different aerosol dilutions. Outliers occur, mainly towards lower delivery.

Aerosol delivery efficiency

Figure 6A shows dilution corrected particle delivery efficiencies, that is, the efficiency with which the particles are sampled from the dilution system into the exposure trumpets, transferred to the exposure chambers and deposited in the PBS samples. Median values range from 0.23% to 0.34%, the first and third quartile span a range of roughly 20% of the median.

Comparison to cigarette smoke

Figure 6B shows dilution corrected nicotine delivery efficiencies measured upon exposure of PBS samples to 3R4F smoke. The direct comparison of the delivery efficiencies of glycerol particles and nicotine in 3R4F smoke (which is predominantly present in the particulate smoke fraction) shows that glycerol particles are delivered with four to tenfold higher efficiency. In contrast to glycerol particles, the delivery efficiency of 3R4F particles is changing with the applied dilution airflow in the Vitrocell®24/48 aerosol exposure system.

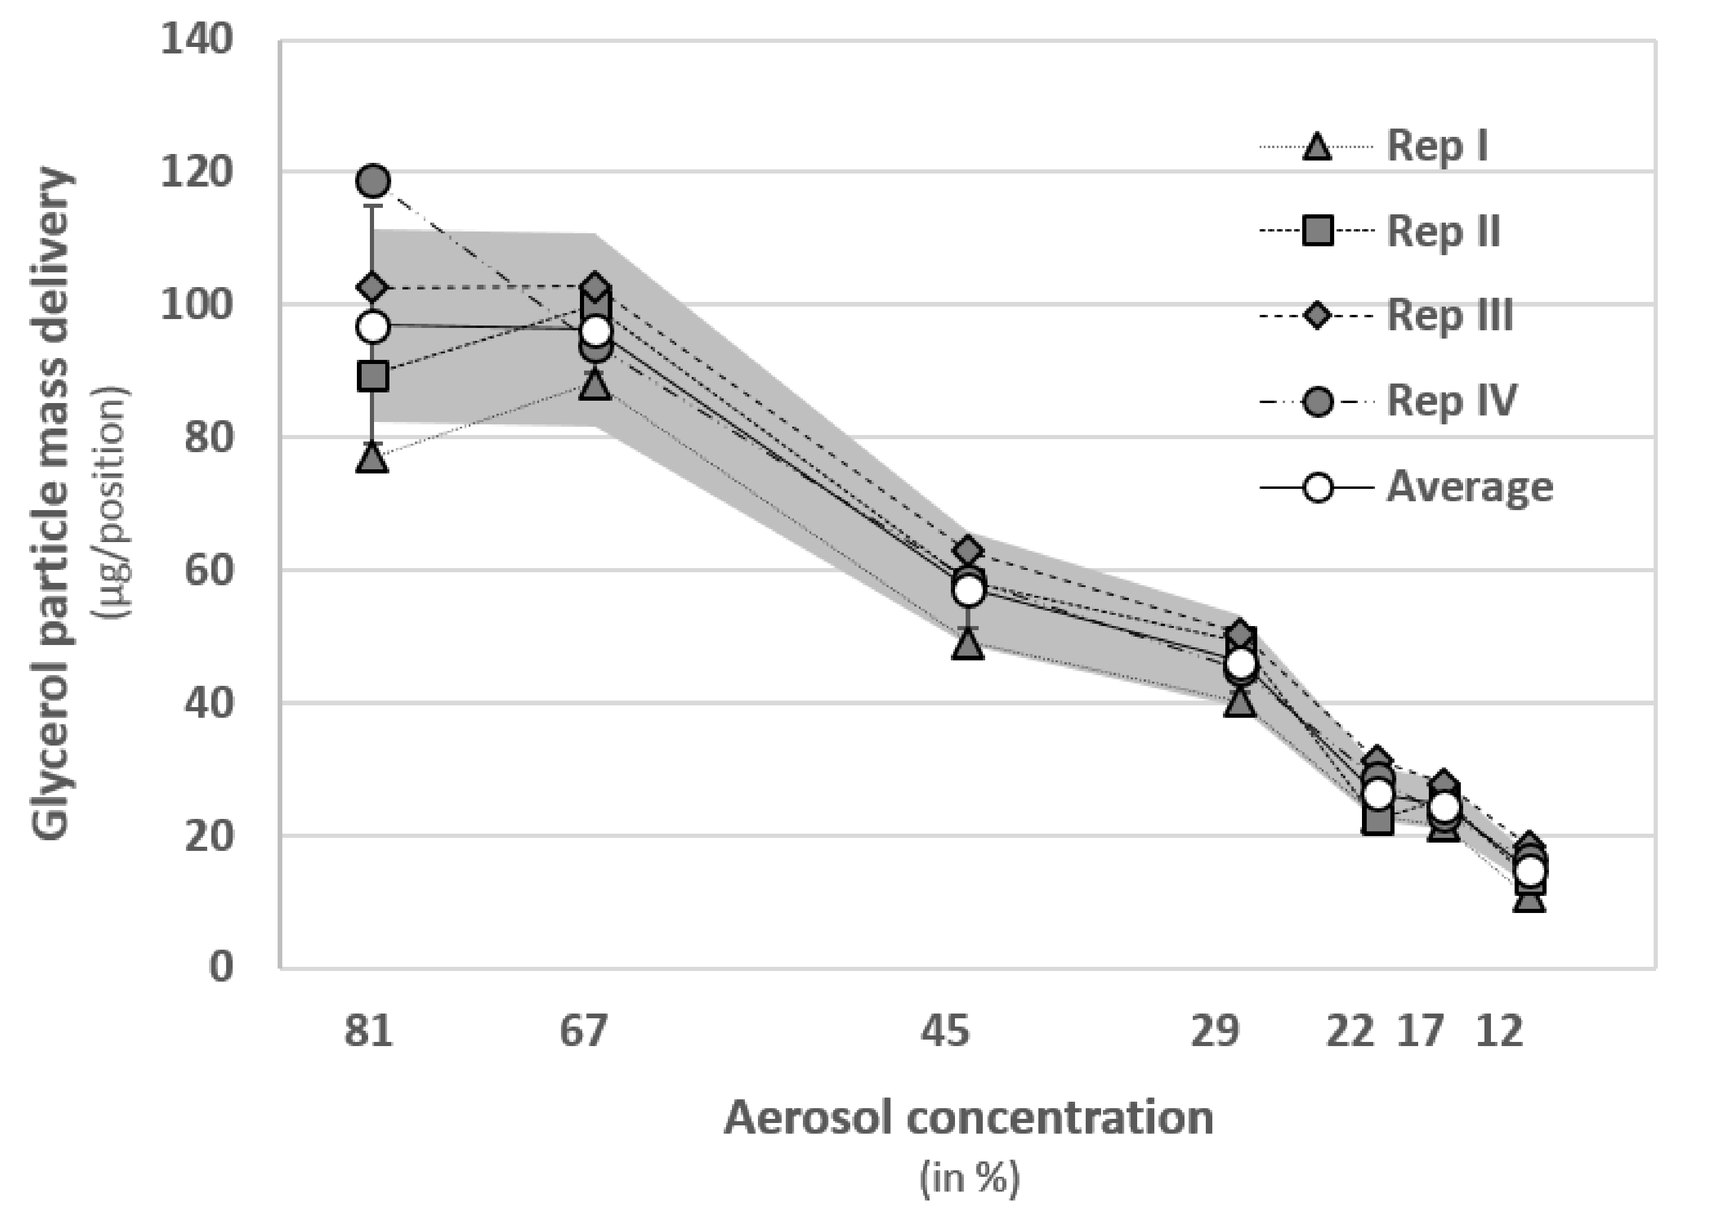
Glycerol particle mass delivery
(µg/position)
Rep I
Rep II
Rep III
Rep IV
Average
Aerosol concentration
(in %)

Figure 4: *Aerosol mass delivery as function of aerosol dilution. Individual exposure runs and the average over four repeated exposures are shown. Error bars represent standard deviations between the four runs. The grey area indicates the average ± 15%.*

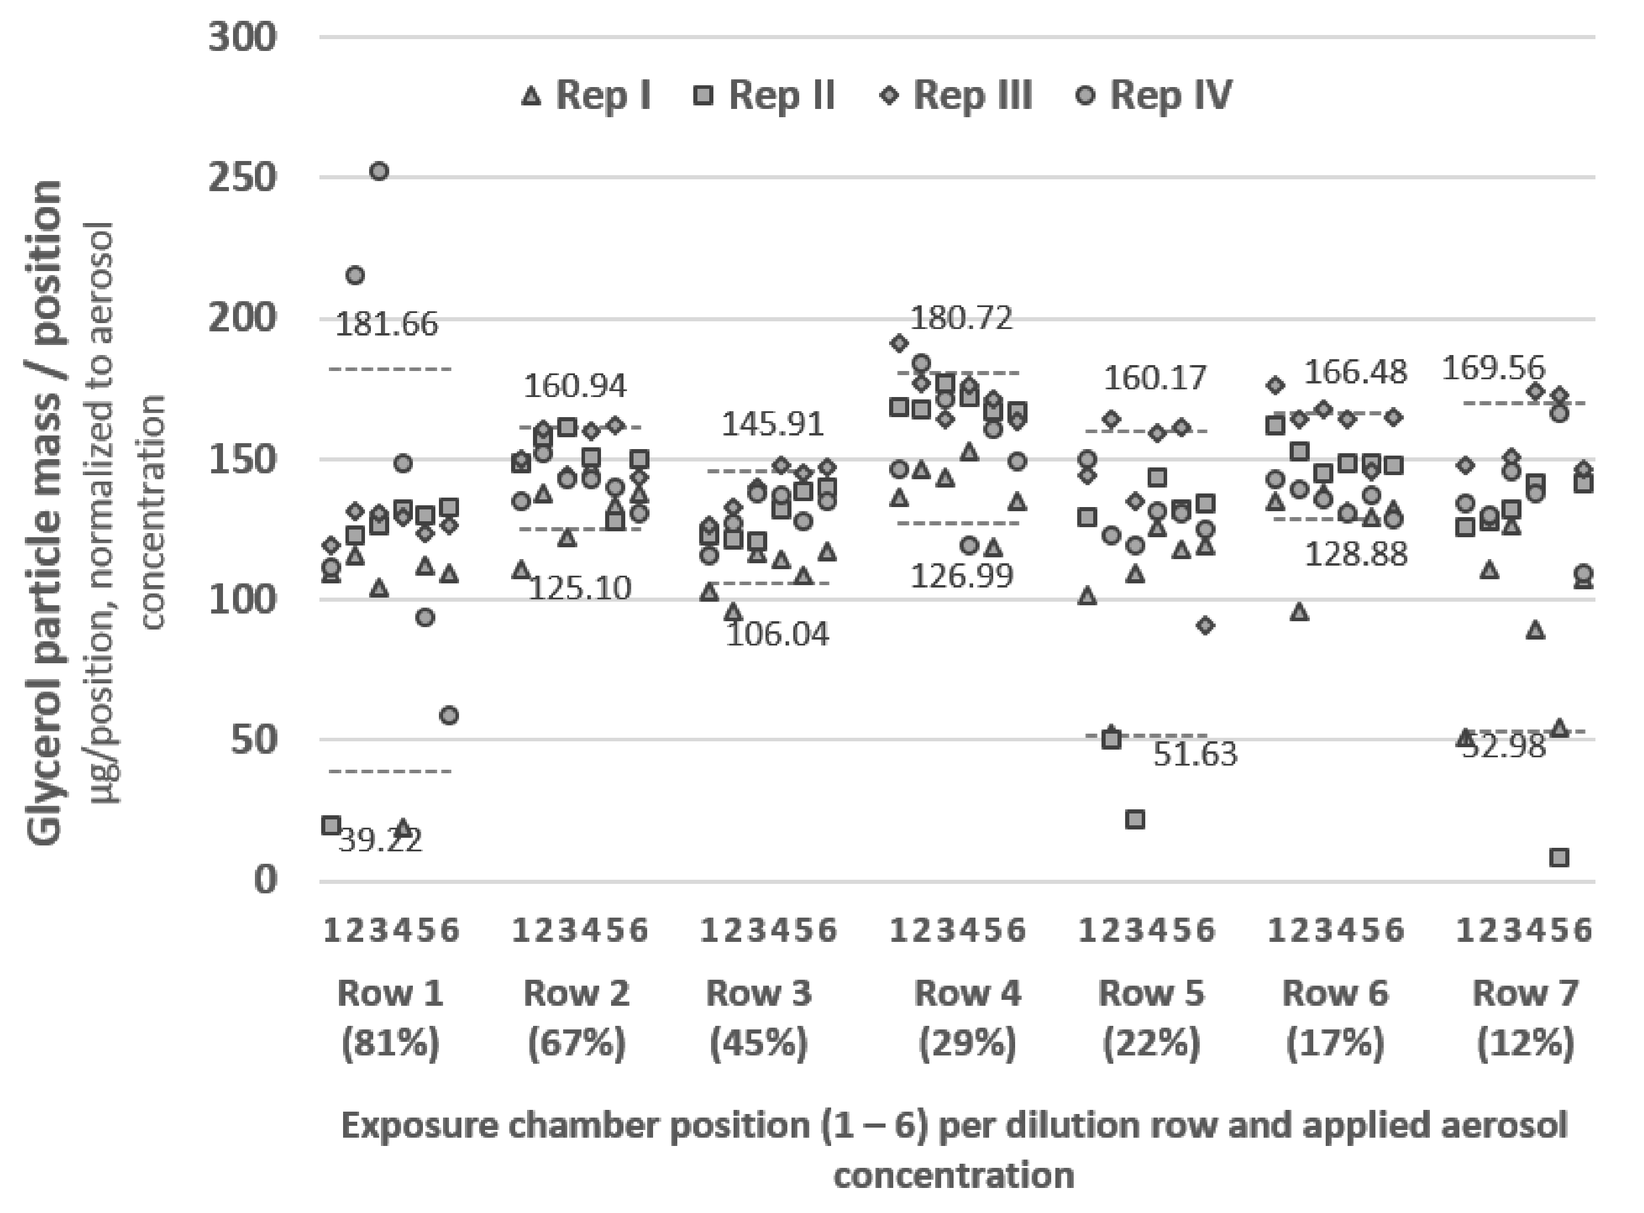
Rep I
Rep II
Rep III
Rep IV
Glycerol particle mass / position
µg/position, normalized to aerosol concentration
181.66
39.22
160.94
125.10
145.91
106.04
180.72
126.99
160.17
51.63
166.48
128.88
169.56
52.98
Row 1 (81%)
Row 2 (67%)
Row 3 (45%)
Row 4 (29%)
Row 5 (22%)
Row 6 (17%)
Row 7 (12%)
Exposure chamber position (1 – 6) per dilution row and applied aerosol concentration

Figure 5: *Aerosol mass delivery per exposure chamber, normalized to the applied aerosol concentration. Dashed lines and numbers indicate the 10st and 90th percentiles.*

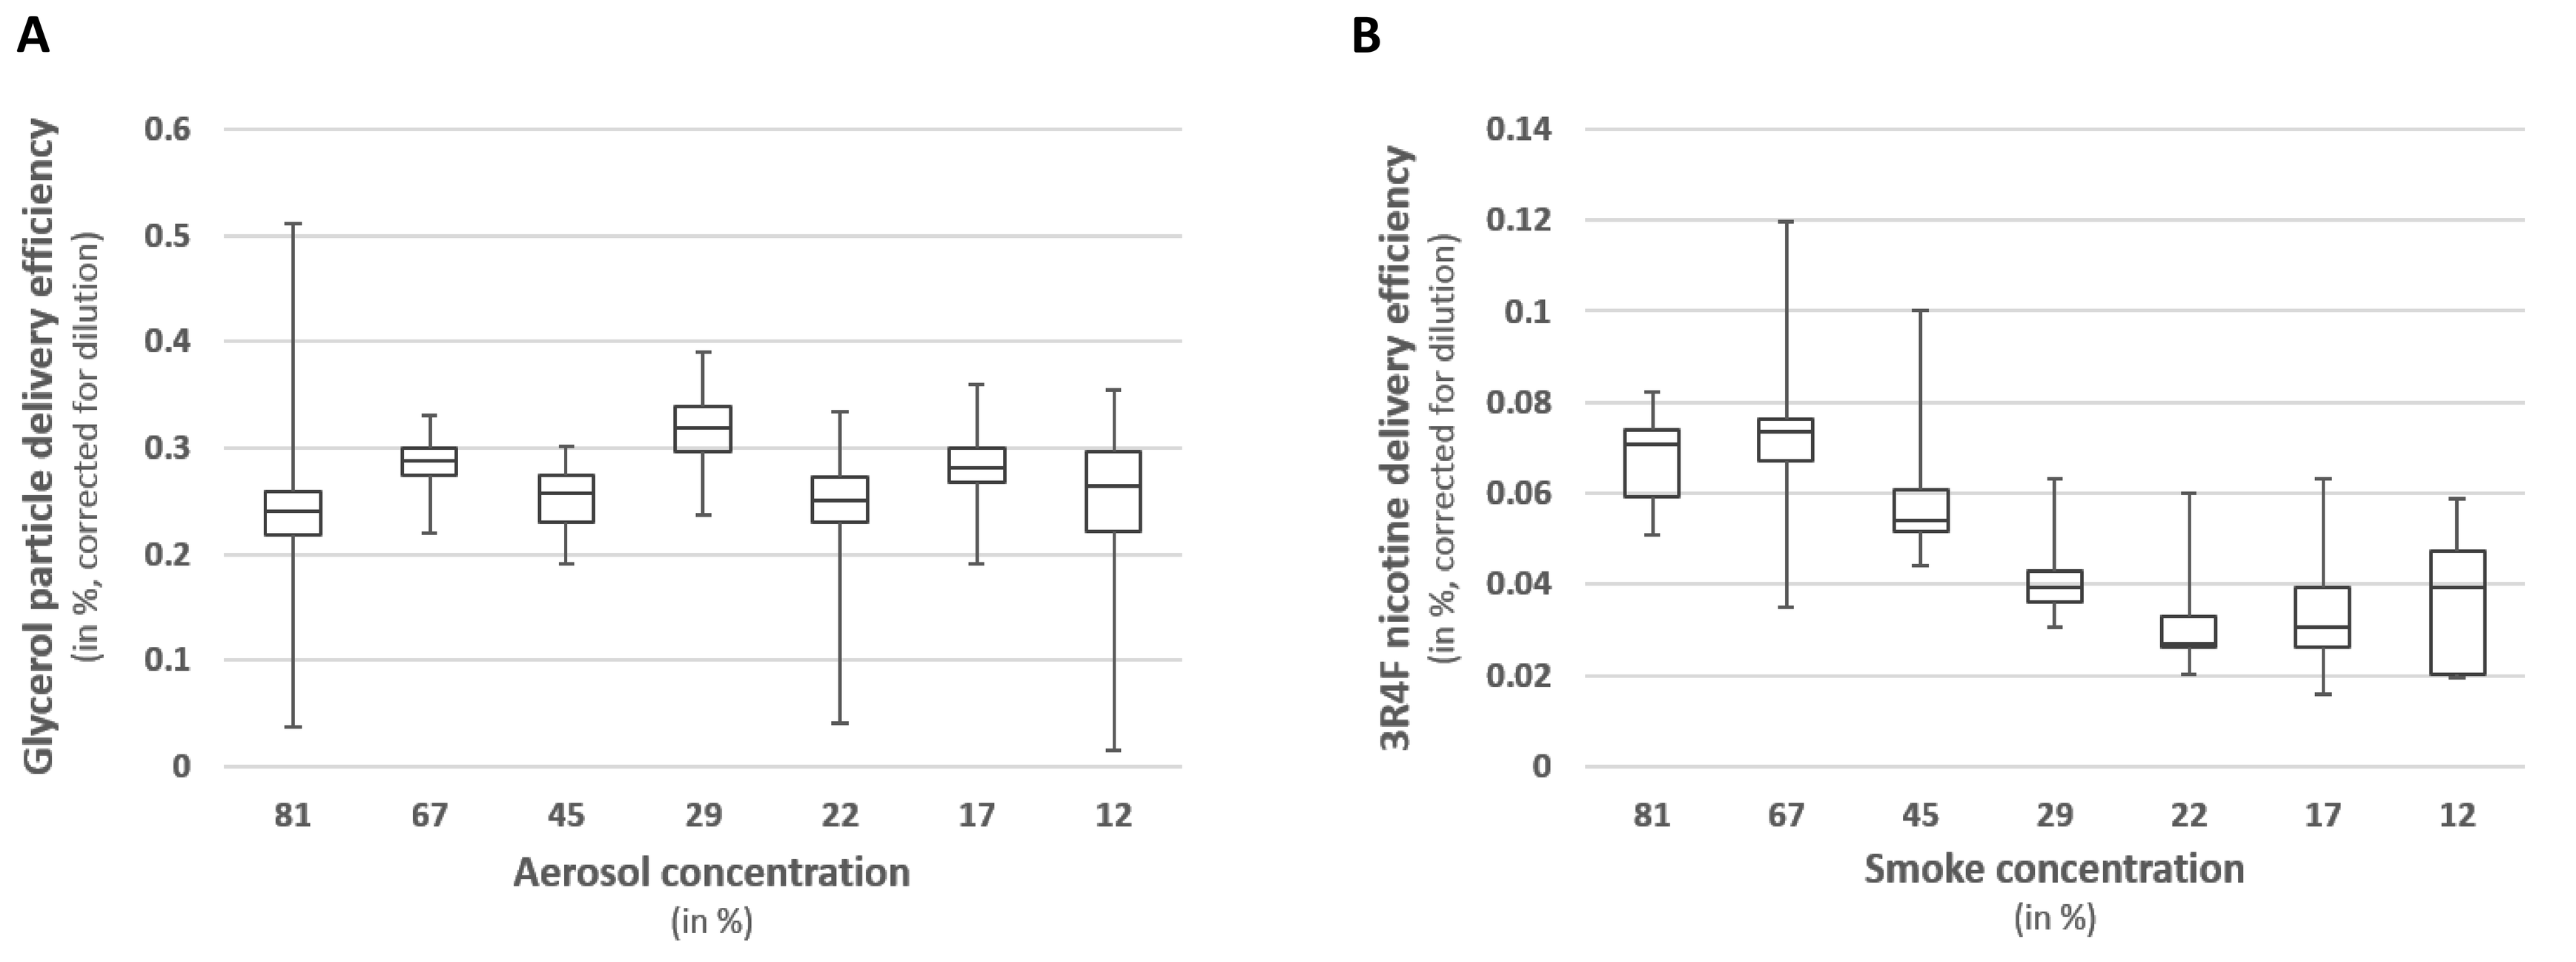
A
Glycerol particle delivery efficiency
(in %, corrected for dilution)
Aerosol concentration
(in %)
B
3R4F nicotine delivery efficiency
(in %, corrected for dilution)
Smoke concentration
(in %)

Figure 6: Dilution corrected delivery efficiency of A) glycerol aerosol and B) 3R4F smoke. Delivery efficiencies were calculated according to Eq 2, the boxplots represent first, second and third quartile, minimal and maximal value.

Discussion and Conclusions

Using a fluorescently labelled glycerol model aerosol, we conducted a characterization of the Vitrocell®24/48 aerosol exposure system for its use in conjunction with liquid aerosols. The choice for the used model aerosol was based on its high reproducibility of generation, its stability and the possibility to include a fluorescent tracer for easy detection of aerosol deposition. The used glycerol aerosol provides a high comparability to aerosols generated by electronic cigarettes or various medical inhalation devices, for instance with respect to the density, viscosity and hydrophobicity of the aerosol material. Our results are therefore representative for these kinds of aerosols and we demonstrate that the Vitrocell®24/48 aerosol exposure is feasible for testing them, given a set of aspects is taken into consideration:

- Particle delivery can be precisely controlled by aerosol dilution, but since full congruency between dilution and delivery is not provided, the ultimately delivered doses need to be determined experimentally.
- The repeatability of exposures is high, if the average particle delivery to the six positions per dilution row is considered.
- Higher variation in the particle delivery was detected on the level of individual exposure chambers.
- The direct comparability between exposures to glycerol aerosols (or aerosols of comparable properties) and exposures to 3R4F smoke, even if exposed under identical exposure systems settings, is not implicitly given and requires experimental confirmation.

References: ***1)*** Steiner et al., Characterization of the Vitrocell® 24/48 aerosol exposure system for its use in exposures to liquid aerosols. Toxicology in Vitro 2017, article in press. ***2)*** Health Canada: Determination of tar, water, nicotine and carbon monoxide in mainstream tobacco smoke. Health Canada Test Method T-115;(1999). ***3)*** Gowadia et al., A transport model for nicotine in the tracheobronchial and pulmonary region of the lung. Inhalation Toxicology, 2010; 22. ***4)*** Majeed et al., Characterization of the Vitrocell® 24/48 in vitro aerosol exposure system using mainstream cigarette smoke. Chemistry Central Journal 2014; 8.

PMI SCIENCE
PHILIP MORRIS INTERNATIONAL

21st Congress of the International Society for Aerosols in Medicine, Santa Fe

03rd - 07th June 2017

Competing Financial Interest

The research described in this poster was funded by Philip Morris International