Comparison of the impacts of an acute exposure to electronic cigarette aerosol and cigarette smoke on small airway epithelial cultures: In vitro systems toxicology assessment

Anita R. Iskandar[1], Athanasios Kondylis[1], Florian Martin[1], Sandro Steiner[1], Shoaib Majeed[1], Patrice Leroy[1], Emmanuel Guedj[1], Celine Merg[1], Keyur Trivedi[1], Stefan Frentzel[1], Nikolai V. Ivanov[1], Utkarsh Doshi[2], K. Monica Lee[2], Willie J. Mckinney Jr[2*], Manuel C. Peitsch[1], Julia Hoeng[1]
[1]PMI R&D, Philip Morris Pro; *Former Altria Client Services employeeducts S.A., Quai Jeanrenaud 5, CH-2000 Neuchâtel, Switzerland; [2]Altria Client Services LLC, Richmond, VA, 23219, USA

Introduction and Objectives

With increasing popularity of electronic cigarettes (ECs), it is important to assess the potential toxicity of EC aerosol exposure. In the context of a harm reduction approach, we examined the effects of whole EC aerosol exposure relative to the effects of mainstream cigarette smoke (CS) exposure using human organotypic small airway cultures. Cultures were exposed at the air-liquid interface to 112 puffs of either undiluted aerosols generated from a MarkTen® cartridge containing various e-liquids (with aerosol formers alone ["Carrier"], with 4% nicotine ["Base"], with 4% nicotine and flavors ["Test Red"] or to diluted CS, in Vitrocell® exposure systems. We conducted a series of independent exposure repetitions to strengthen the accuracy of the observation. Concentrations of the deposited nicotine and carbonyls in the exposure chamber were measured as markers of exposure. Biological endpoints investigated included histology, cytotoxicity, inflammatory mediators, and gene microarray. Alterations in morphology or cytotoxicity were not observed in small airway cultures exposed to undiluted EC aerosols despite resulting in higher nicotine deposition in the chamber than that found following CS exposure. Increased loss of cilia was recorded in cultures exposed to CS but not in cultures exposed to any EC aerosol. Media of cultures exposed to CS had generally greater levels of inflammatory mediators in comparison to cultures exposed to EC aerosols. CS exposure also elicited a greater number of differentially expressed genes. Based on a network-based enrichment analysis, the transcriptome data showed that the exposures impacted different cellular processes (e.g., cell fate, proliferation, stress, and inflammatory response) with greater impacts following CS exposure than following EC aerosol exposure. These collective endpoints demonstrated that EC aerosols had significantly lower biological impact in small airway cultures, in comparison to CS exposure.

Methods

Experimental Phase

1 2 3

● independent smoke/aerosol generation

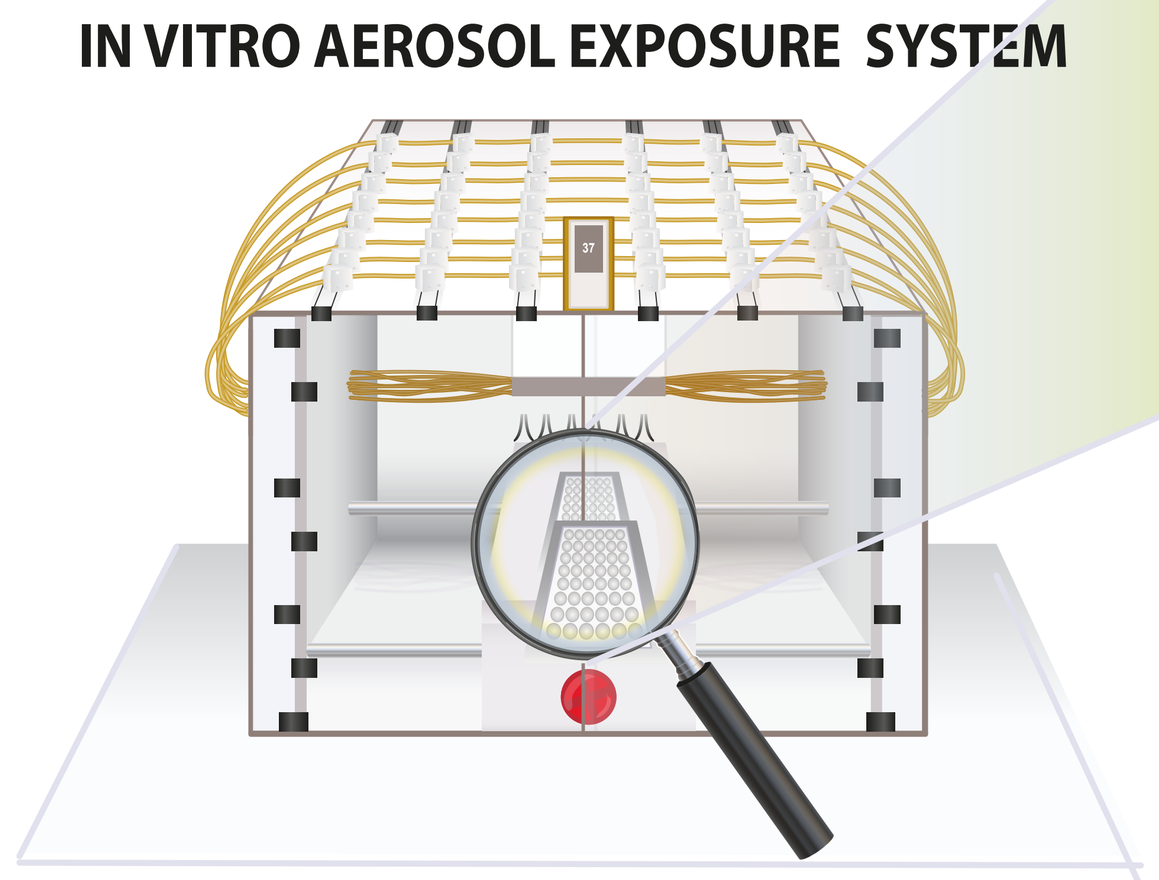
IN VITRO AEROSOL EXPOSURE SYSTEM

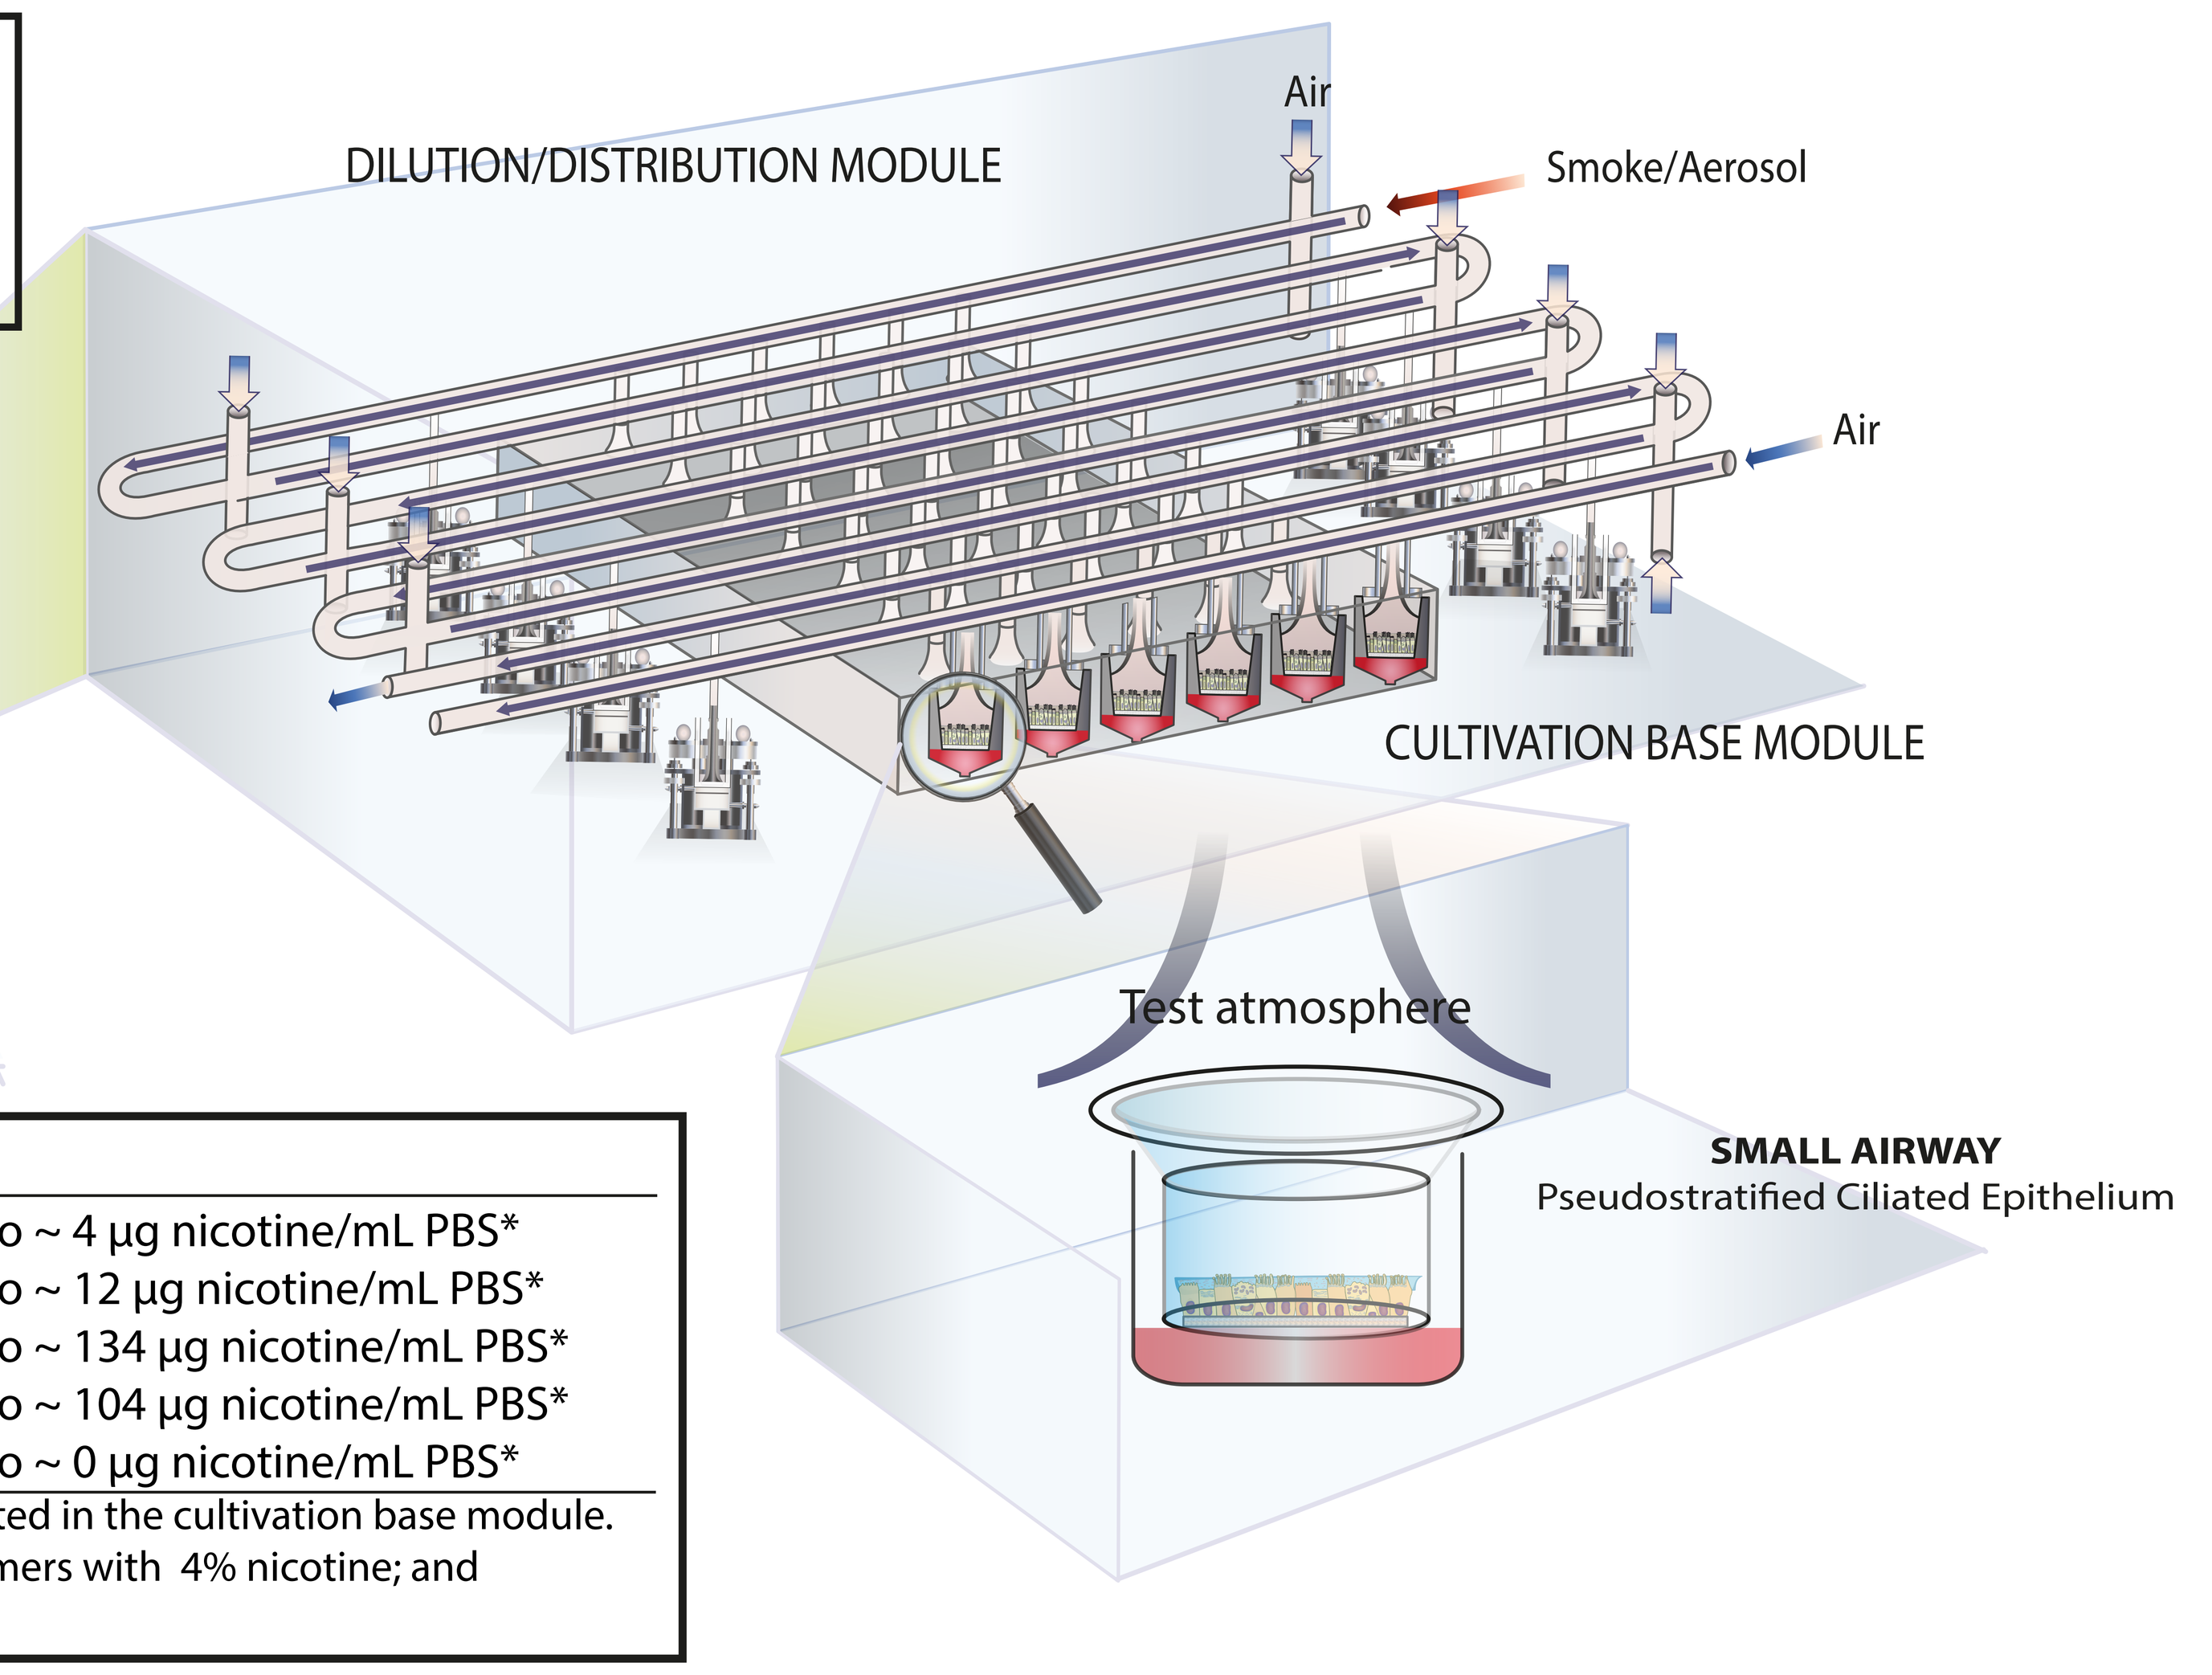
DILUTION/DISTRIBUTION MODULE
Air
Smoke/Aerosol
Air
CULTIVATION BASE MODULE
Test atmosphere
SMALL AIRWAY
Pseudostratified Ciliated Epithelium

Dose of Exposure

7% 3R4F CS for 28 min	equivalent to ~ 4 µg nicotine/mL PBS*
13% 3R4F CS for 28 min	equivalent to ~ 12 µg nicotine/mL PBS*
100% Test Red EC aerosol for 28 min	equivalent to ~ 134 µg nicotine/mL PBS*
100% Base EC aerosol for 28 min	equivalent to ~ 104 µg nicotine/mL PBS*
100% Carrier EC aerosol for 28 min	equivalent to ~ 0 µg nicotine/mL PBS*

*Nicotine was measured in phosphate-buffered saline (PBS) located in the cultivation base module. Carrier contains aerosol formers alone; Base contains aerosol formers with 4% nicotine; and Test Red contains aerosol formers with 4% nicotine and flavors.

ENDPOINTS

	Time Point of Measurement				
	Pre-Exposure	Post-Exposure			
Determined using exposed PBS samples		0 h	2 h	24 h	48 h
Deposited nicotine in the exposure chamber	-	✓	-	-	-
Determined using the epithelial cultures					
Culture histology	-	-	-	-	✓
Ciliary beating frequency	✓	✓	-	✓	✓
Transcriptomics	-	-	✓	✓	-
Determined using the basolateral medium samples					
Secreted inflammatory mediators	-	-	-	-	✓

The impacts of an acute 28-minute exposure to CS (from the 3R4F reference cigarette, University of Kentucky) and to EC aerosols (from Test Red, Base, and Carrier formulations; Altria Client Services LLC) were assessed using human organotypic small airway epithelial cultures (reconstituted from the small airway epithelial cells of 72 year-old male, nonsmoker donor). A paired design was implemented: in parallel to the exposure to CS or e-vapor aerosol, cultures were also exposed to air in the same exposure module. For each of the culture models, a series of nine experimental runs was conducted to increase the assessment robustness. For the histological analysis, the cross-sections of the organotypic epithelium cultures were analyzed after hematoxylin and eosin (H&E) and Alcian blue staining. Ciliary beating measurement was conducted using the Sisson Ammons Video Analysis system on a total of 512 video frames recorded from the center of the insert surface. Concentrations of inflammatory mediators were measured from the basolateral medium of the exposed cultures using Luminex® xMAP® technology and commercially available assay panels (EMD Millipore Corp) according to the manufacturer's instructions. Messenger RNA microarrays were done using 100 ng of total RNA (per sample) that were reverse-transcribed and amplified to cRNA using the Affymetrix® HT 3' IVT PLUS kit.

Results

Culture Morphology Following Exposure

13% 3R4F CS
for 28 min (112 puffs)
(12 µg nicotine/mL PBS)

100% Test Red EC aerosol
for 28 min (112 puffs)
(134 µg nicotine/mL PBS)

100% Base EC aerosol
for 28 min (112 puffs)
(104 µg nicotine/mL PBS)

100% Carrier EC aerosol
for 28 min (112 puffs)
(0 µg nicotine/mL PBS)

100% Air
for 28 min (112 puffs)
(Control)

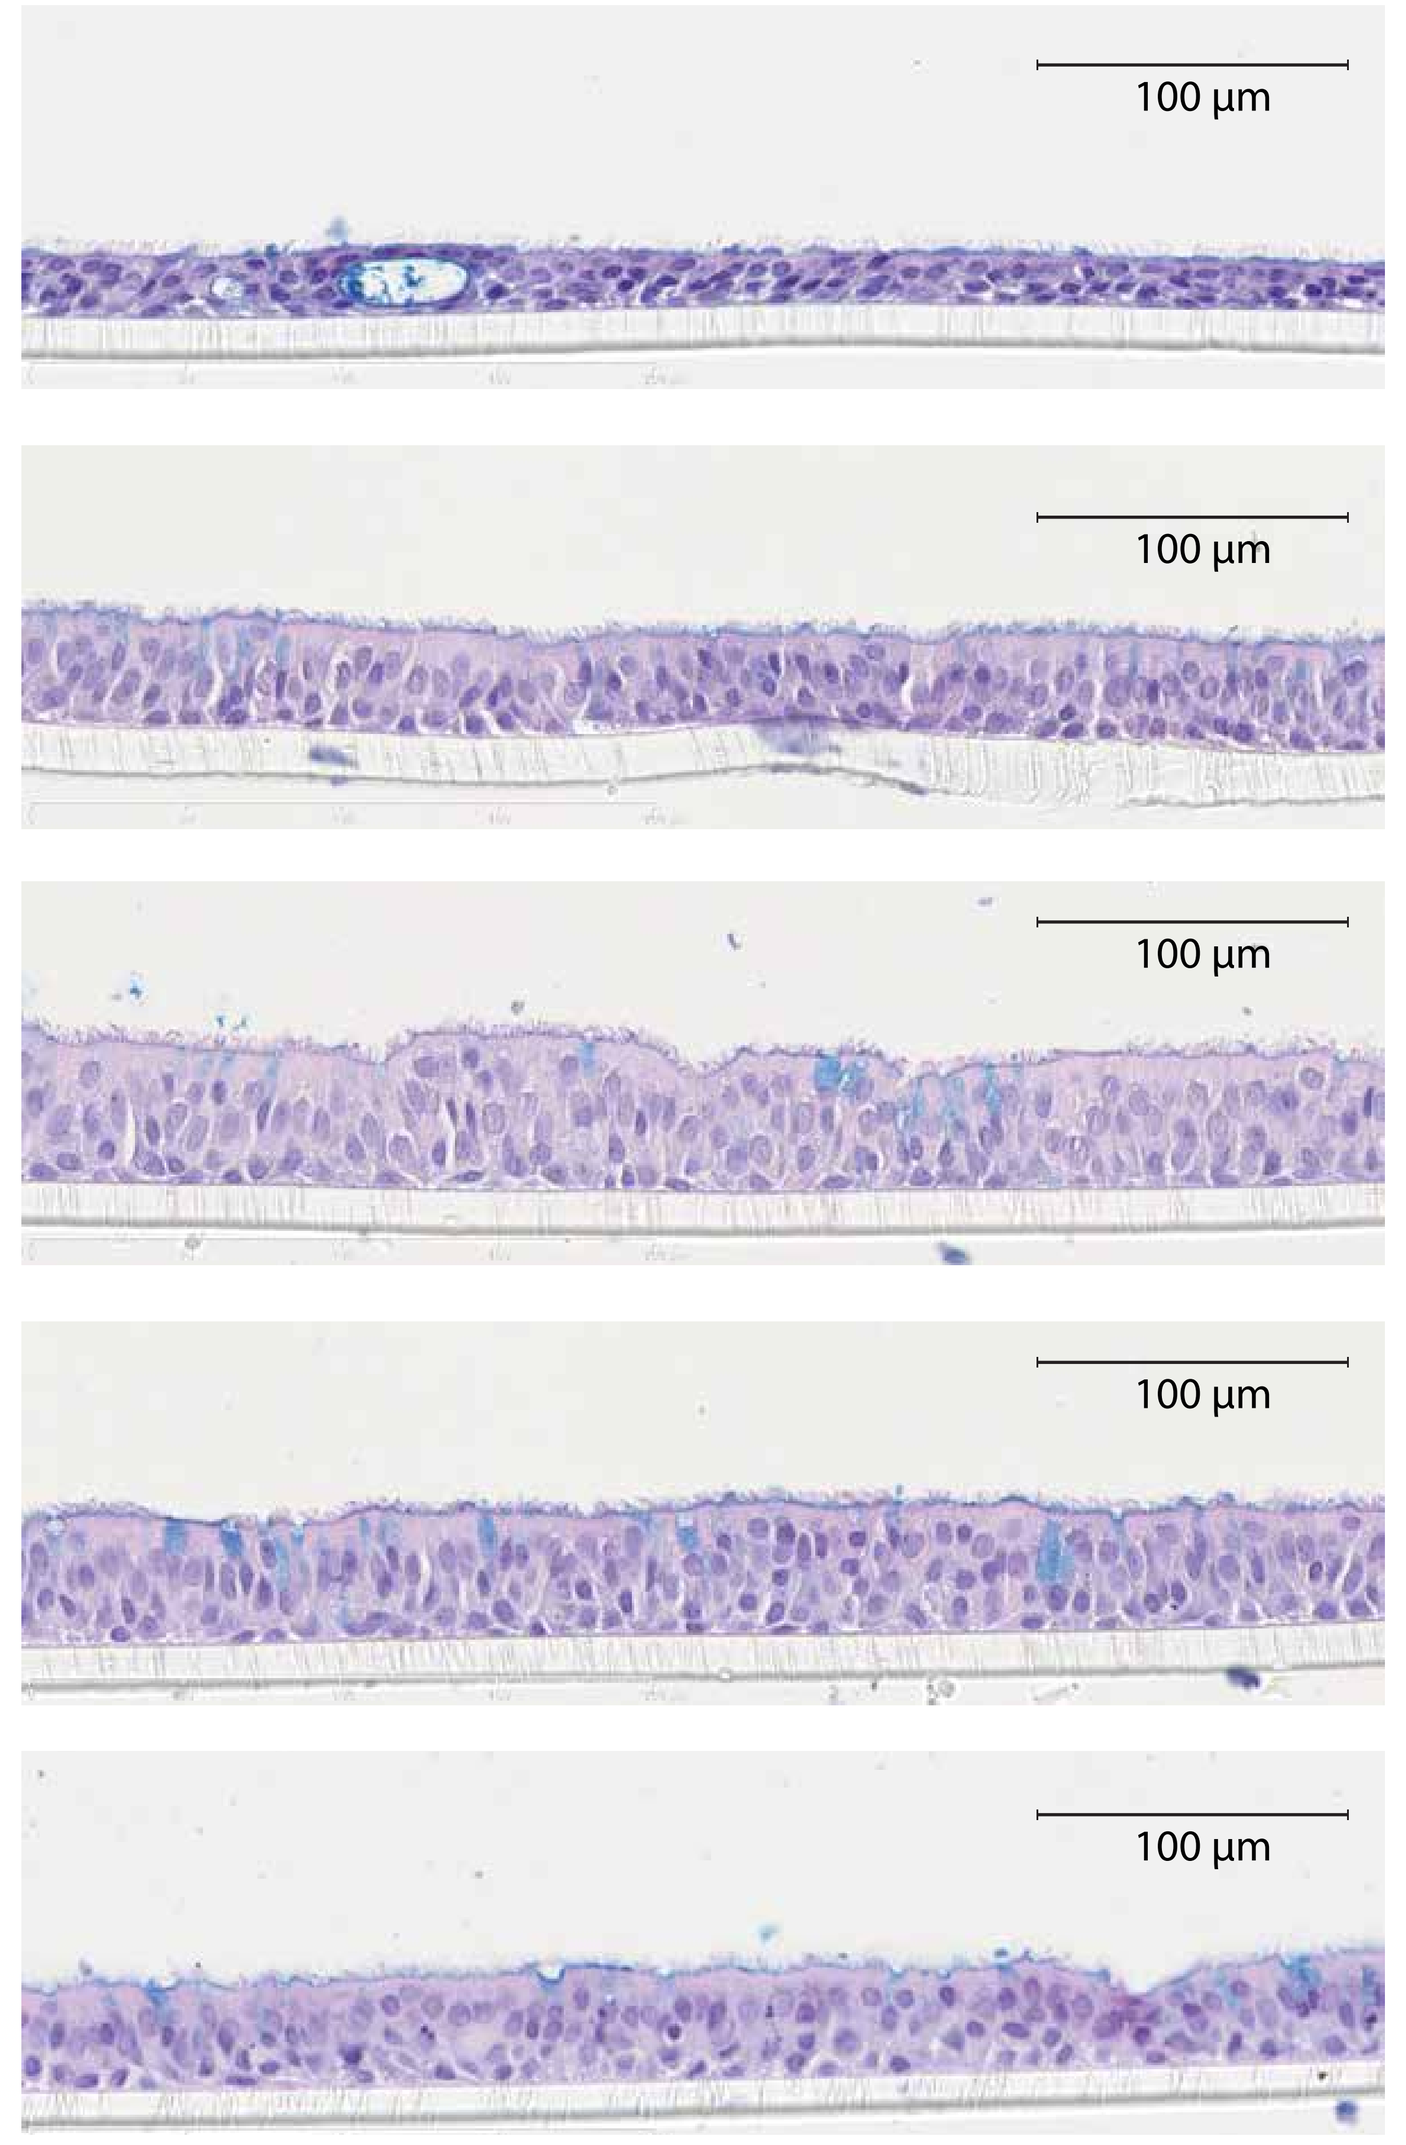
100 µm

Representative images of the H&E-and Alcian blue-stained sections are shown. In small airway cultures, thinning of the epithelium was seen in following exposure to the 28-minute (112-puff) 13% 3R4F CS, corresponding to around 12 µg/mL deposited nicotine. In contrast, exposure to undiluted Test Red or Base aerosols did not alter the culture morphology despite delivering higher nicotine concentrations.

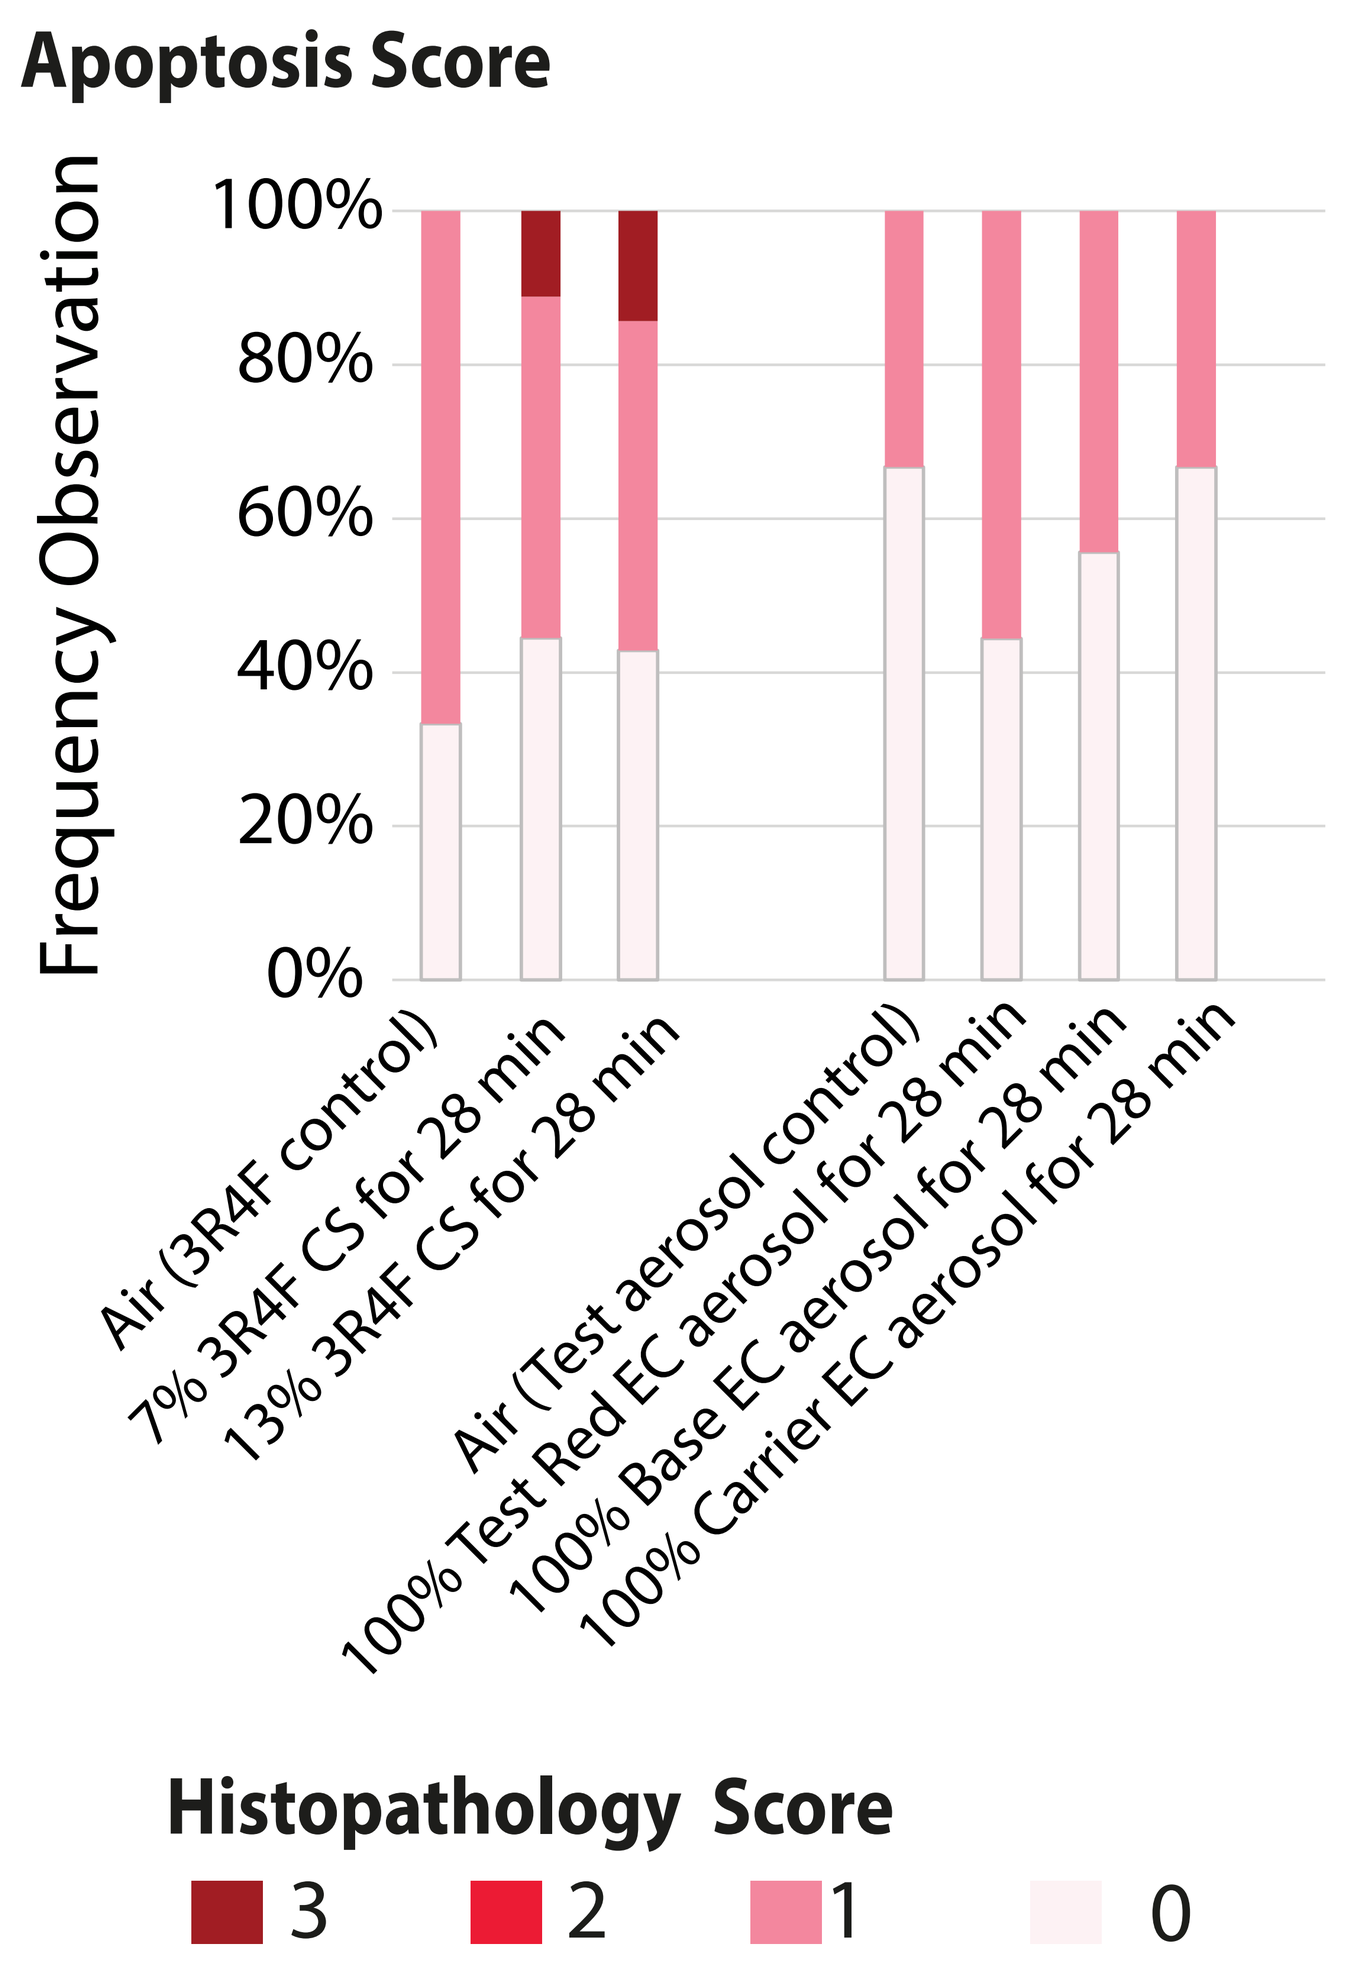
Apoptosis Score
Frequency Observation
100%
80%
60%
40%
20%
0%
Air (3R4F control)
7% 3R4F CS for 28 min
13% 3R4F CS for 28 min
Air (Test aerosol control)
100% Test Red EC aerosol for 28 min
100% Base EC aerosol for 28 min
100% Carrier EC aerosol for 28 min
Histopathology Score
3
2
1
0

Increased apoptosis was detected following exposure to 7% or 13% 3R4F CS for 28 min (112 puffs) compared with the exposure to air. Conversely, it was not detected following exposure to 100% Test Red, Base, or Carrier aerosols for the same duration.

Ciliary Beating Frequency

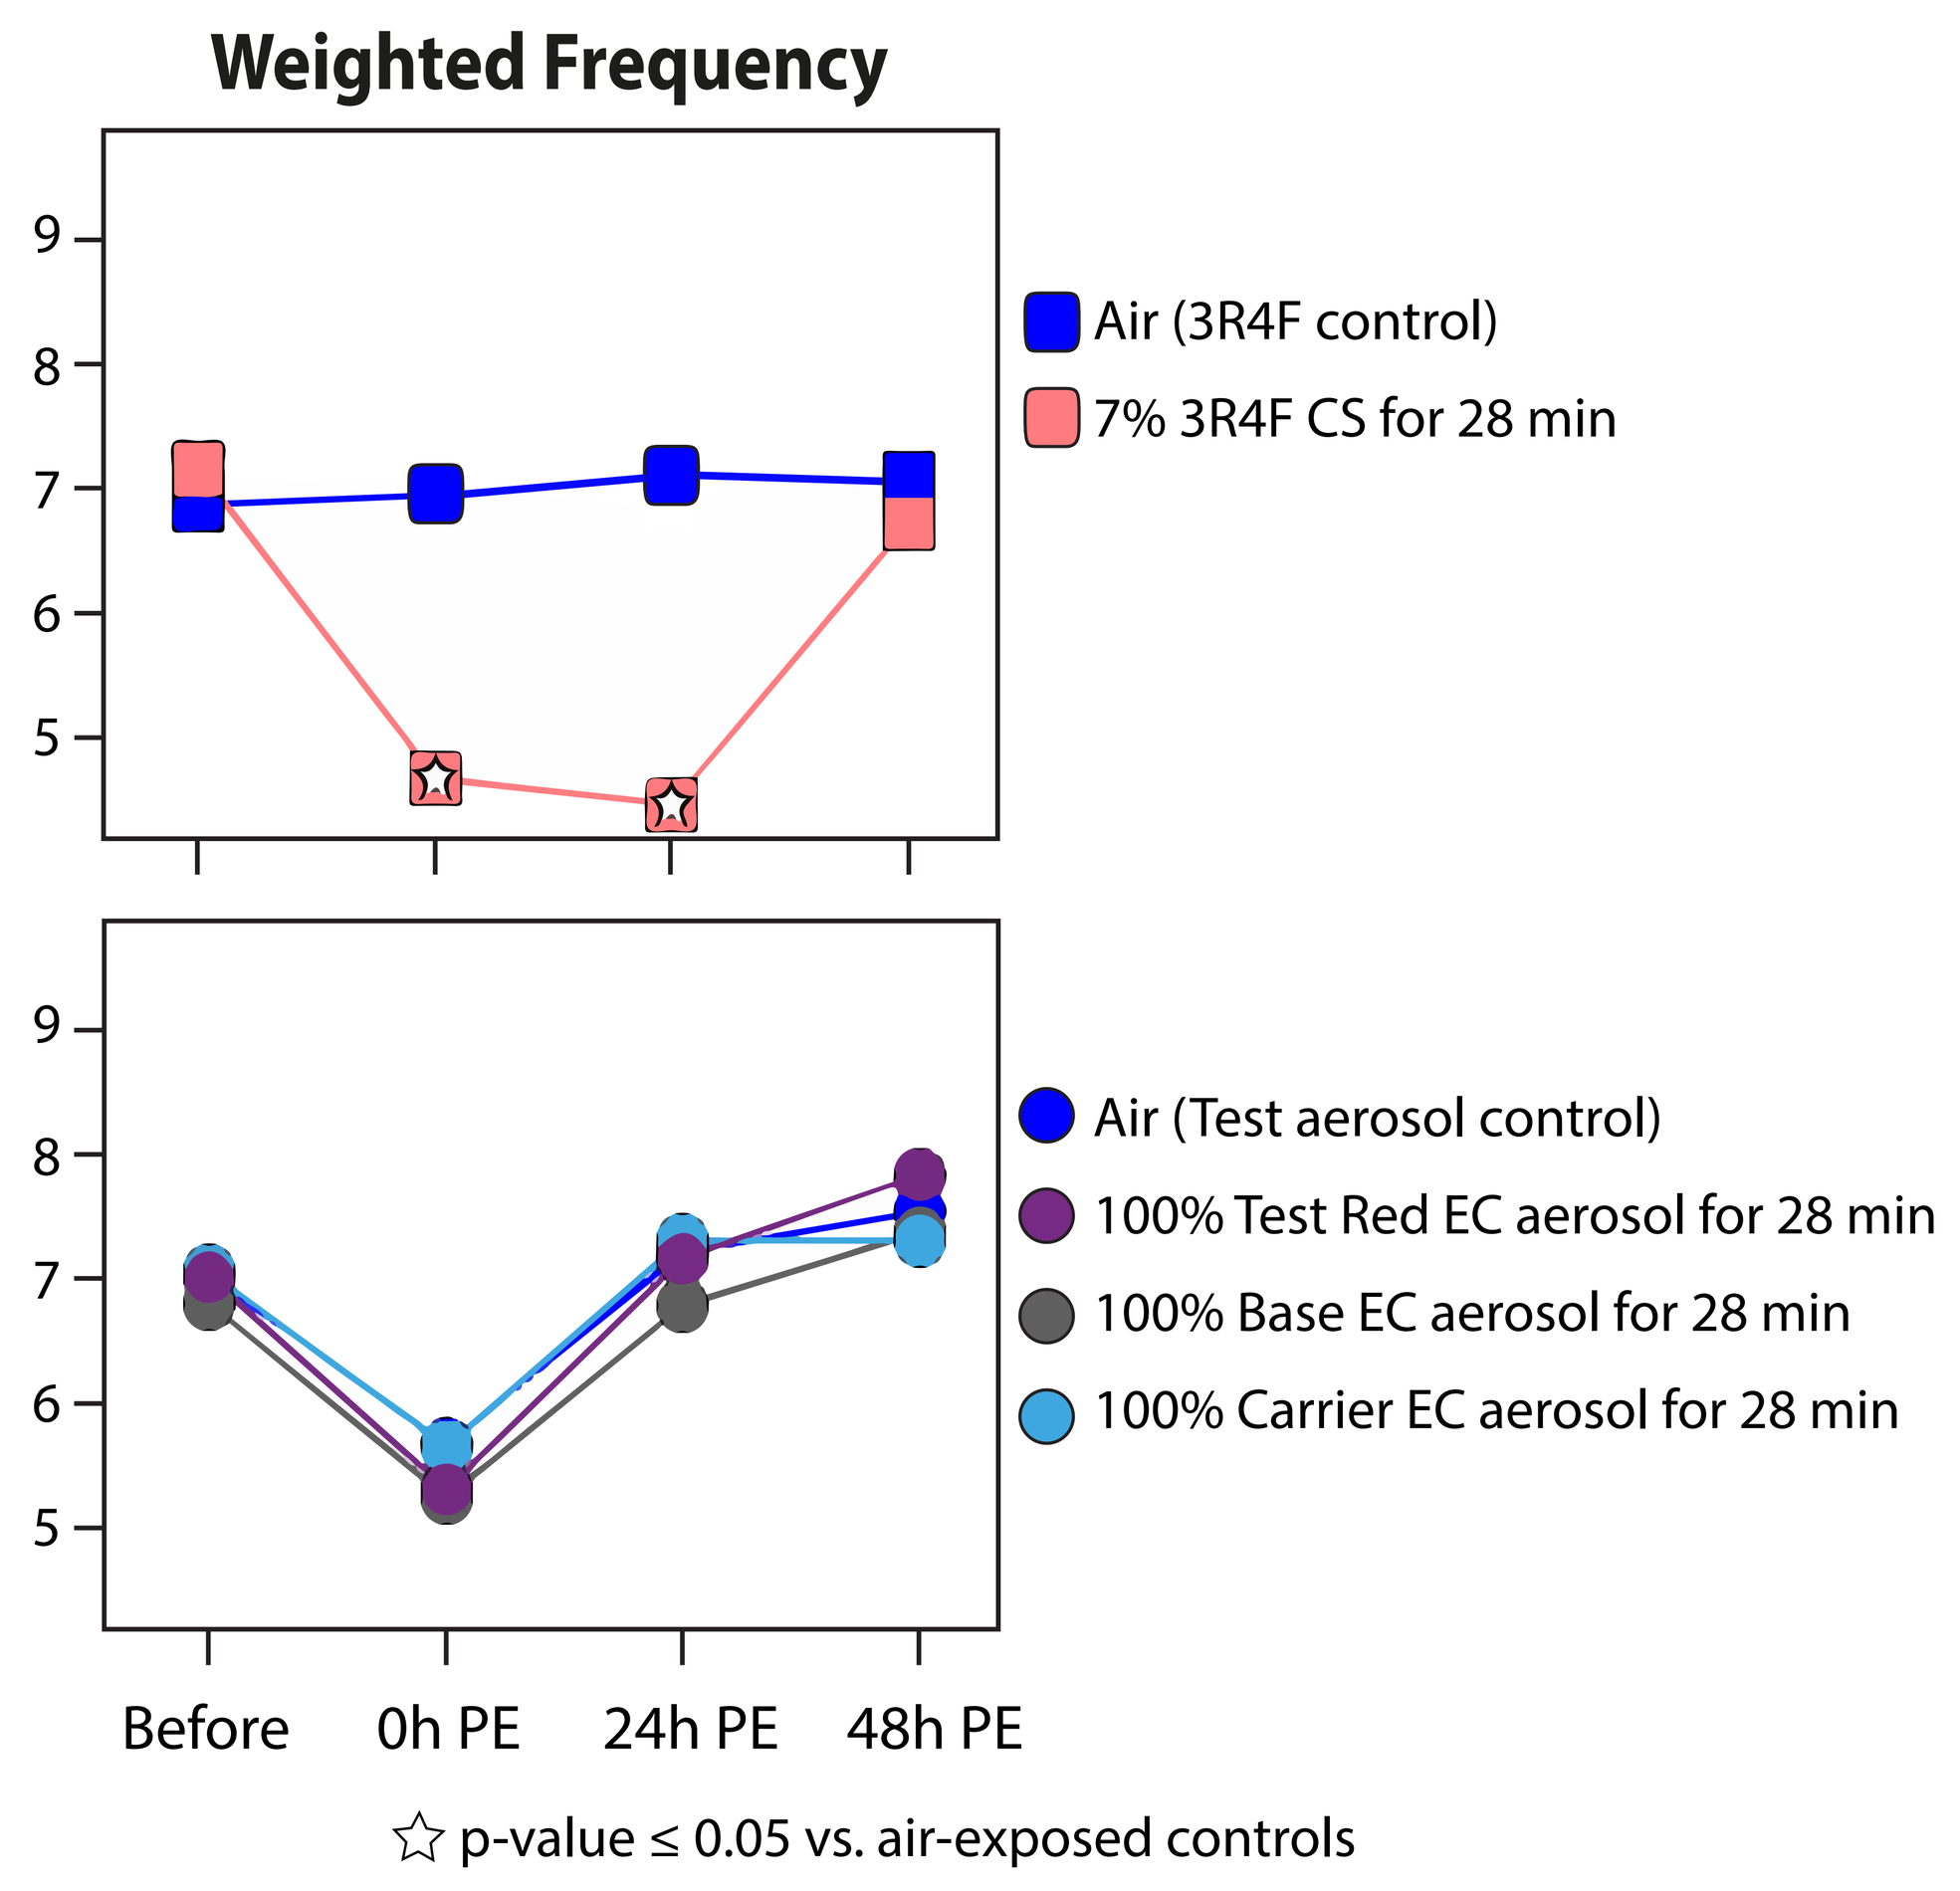
Weighted Frequency
Air (3R4F control)
7% 3R4F CS for 28 min
Air (Test aerosol control)
100% Test Red EC aerosol for 28 min
100% Base EC aerosol for 28 min
100% Carrier EC aerosol for 28 min
Before
0h PE
24h PE
48h PE
☆ p-value ≤ 0.05 vs. air-exposed controls

Reduced ciliary beating frequencies were detected following exposure to 7% 3R4F CS for 28 minutes (112 puffs) compared with exposure to air. The reduced frequencies remained detected at 24 hours post-exposure and finally recovered at 48 hours post-exposure. Exposure to 100% Test Red, Base, or Carrier aerosols, as well as the air-exposed controls, for the same duration resulted in a slight reduction in ciliary beating frequencies; however, this was not statistically significant and recovered at the 24 hours post-exposure time point.

Network Enrichment Based on the Transcriptome Changes

7% 3R4F CS
for 28 min (112 puffs)
(4 µg nicotine/mL PBS)

100% Test Red EC aerosol
for 28 min (112 puffs)
(134 µg nicotine/mL PBS)

100% Base EC aerosol
for 28 min (112 puffs)
(104 µg nicotine/mL PBS)

100% Carrier EC aerosl
for 28 min (112 puffs)
(0 µg nicotine/mL PBS)

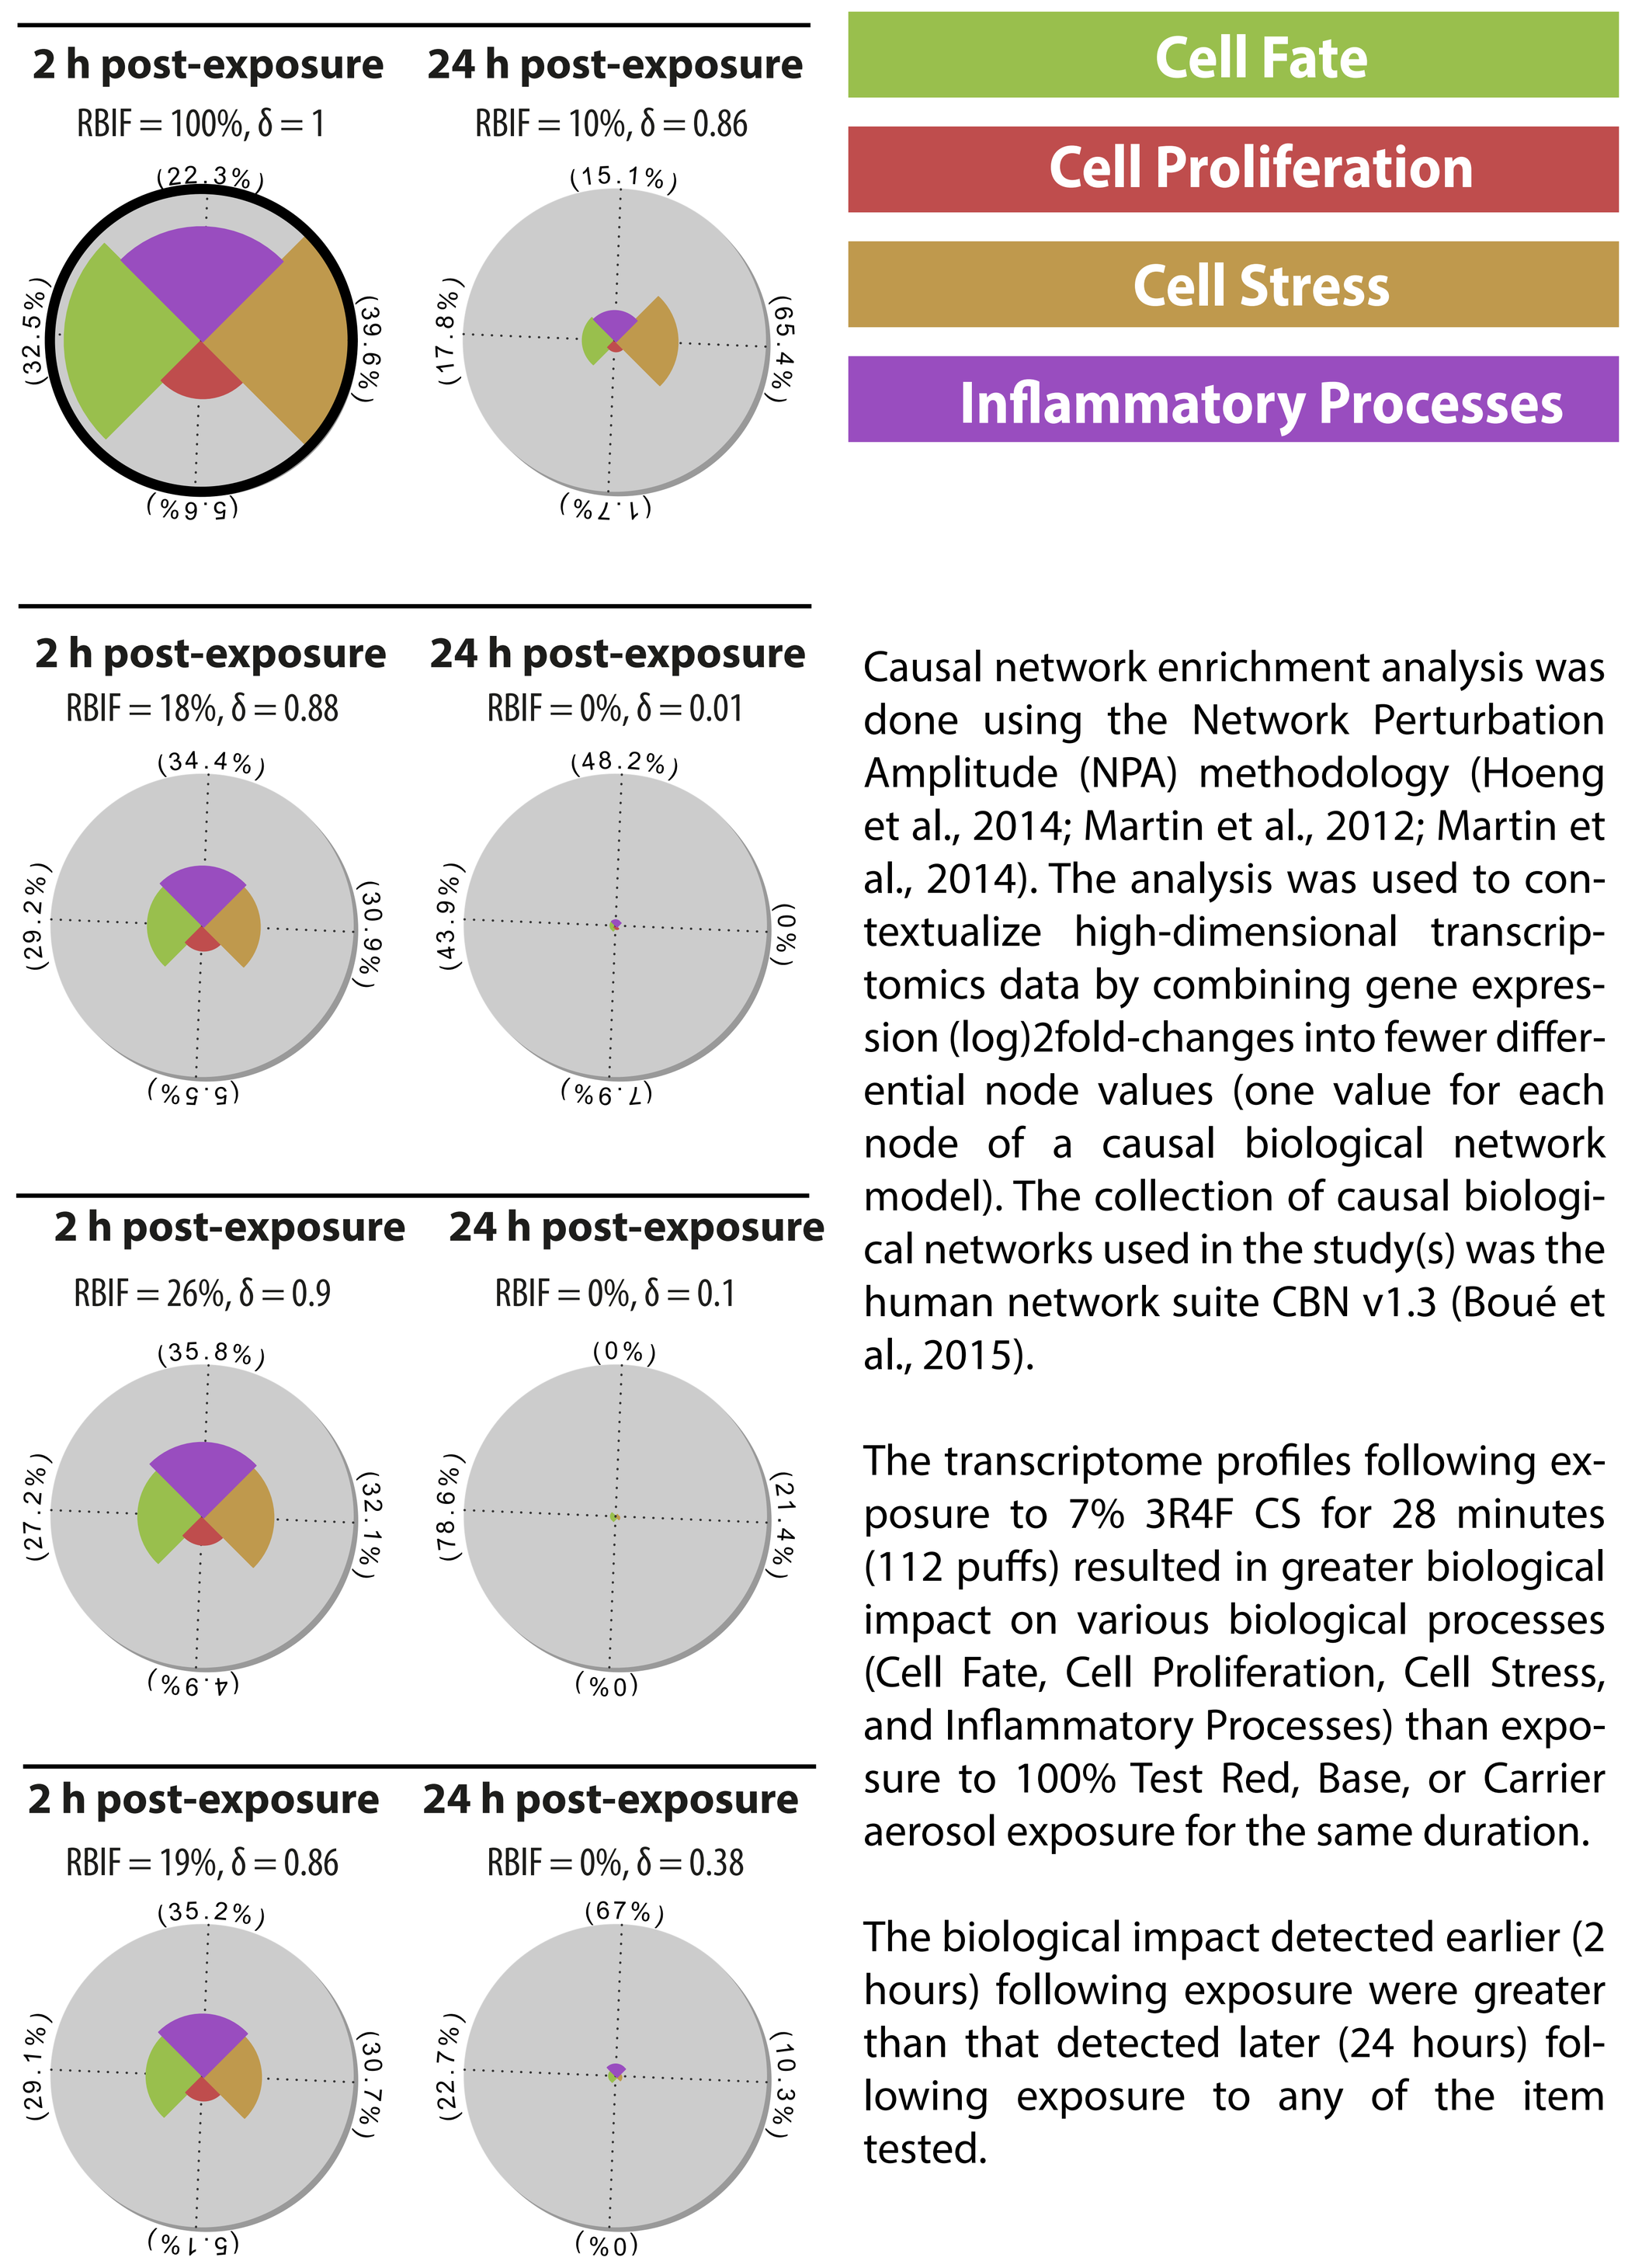
2 h post-exposure
24 h post-exposure
RBIF = 100%, δ = 1
RBIF = 10%, δ = 0.86
RBIF = 18%, δ = 0.88
RBIF = 0%, δ = 0.01
RBIF = 26%, δ = 0.9
RBIF = 0%, δ = 0.1
RBIF = 19%, δ = 0.86
RBIF = 0%, δ = 0.38
Cell Fate
Cell Proliferation
Cell Stress
Inflammatory Processes

Causal network enrichment analysis was done using the Network Perturbation Amplitude (NPA) methodology (Hoeng et al., 2014; Martin et al., 2012; Martin et al., 2014). The analysis was used to contextualize high-dimensional transcriptomics data by combining gene expression (log)2fold-changes into fewer differential node values (one value for each node of a causal biological network model). The collection of causal biological networks used in the study(s) was the human network suite CBN v1.3 (Boué et al., 2015).

The transcriptome profiles following exposure to 7% 3R4F CS for 28 minutes (112 puffs) resulted in greater biological impact on various biological processes (Cell Fate, Cell Proliferation, Cell Stress, and Inflammatory Processes) than exposure to 100% Test Red, Base, or Carrier aerosol exposure for the same duration.

The biological impact detected earlier (2 hours) following exposure were greater than that detected later (24 hours) following exposure to any of the item tested.

Secretion of Inflammatory Mediators

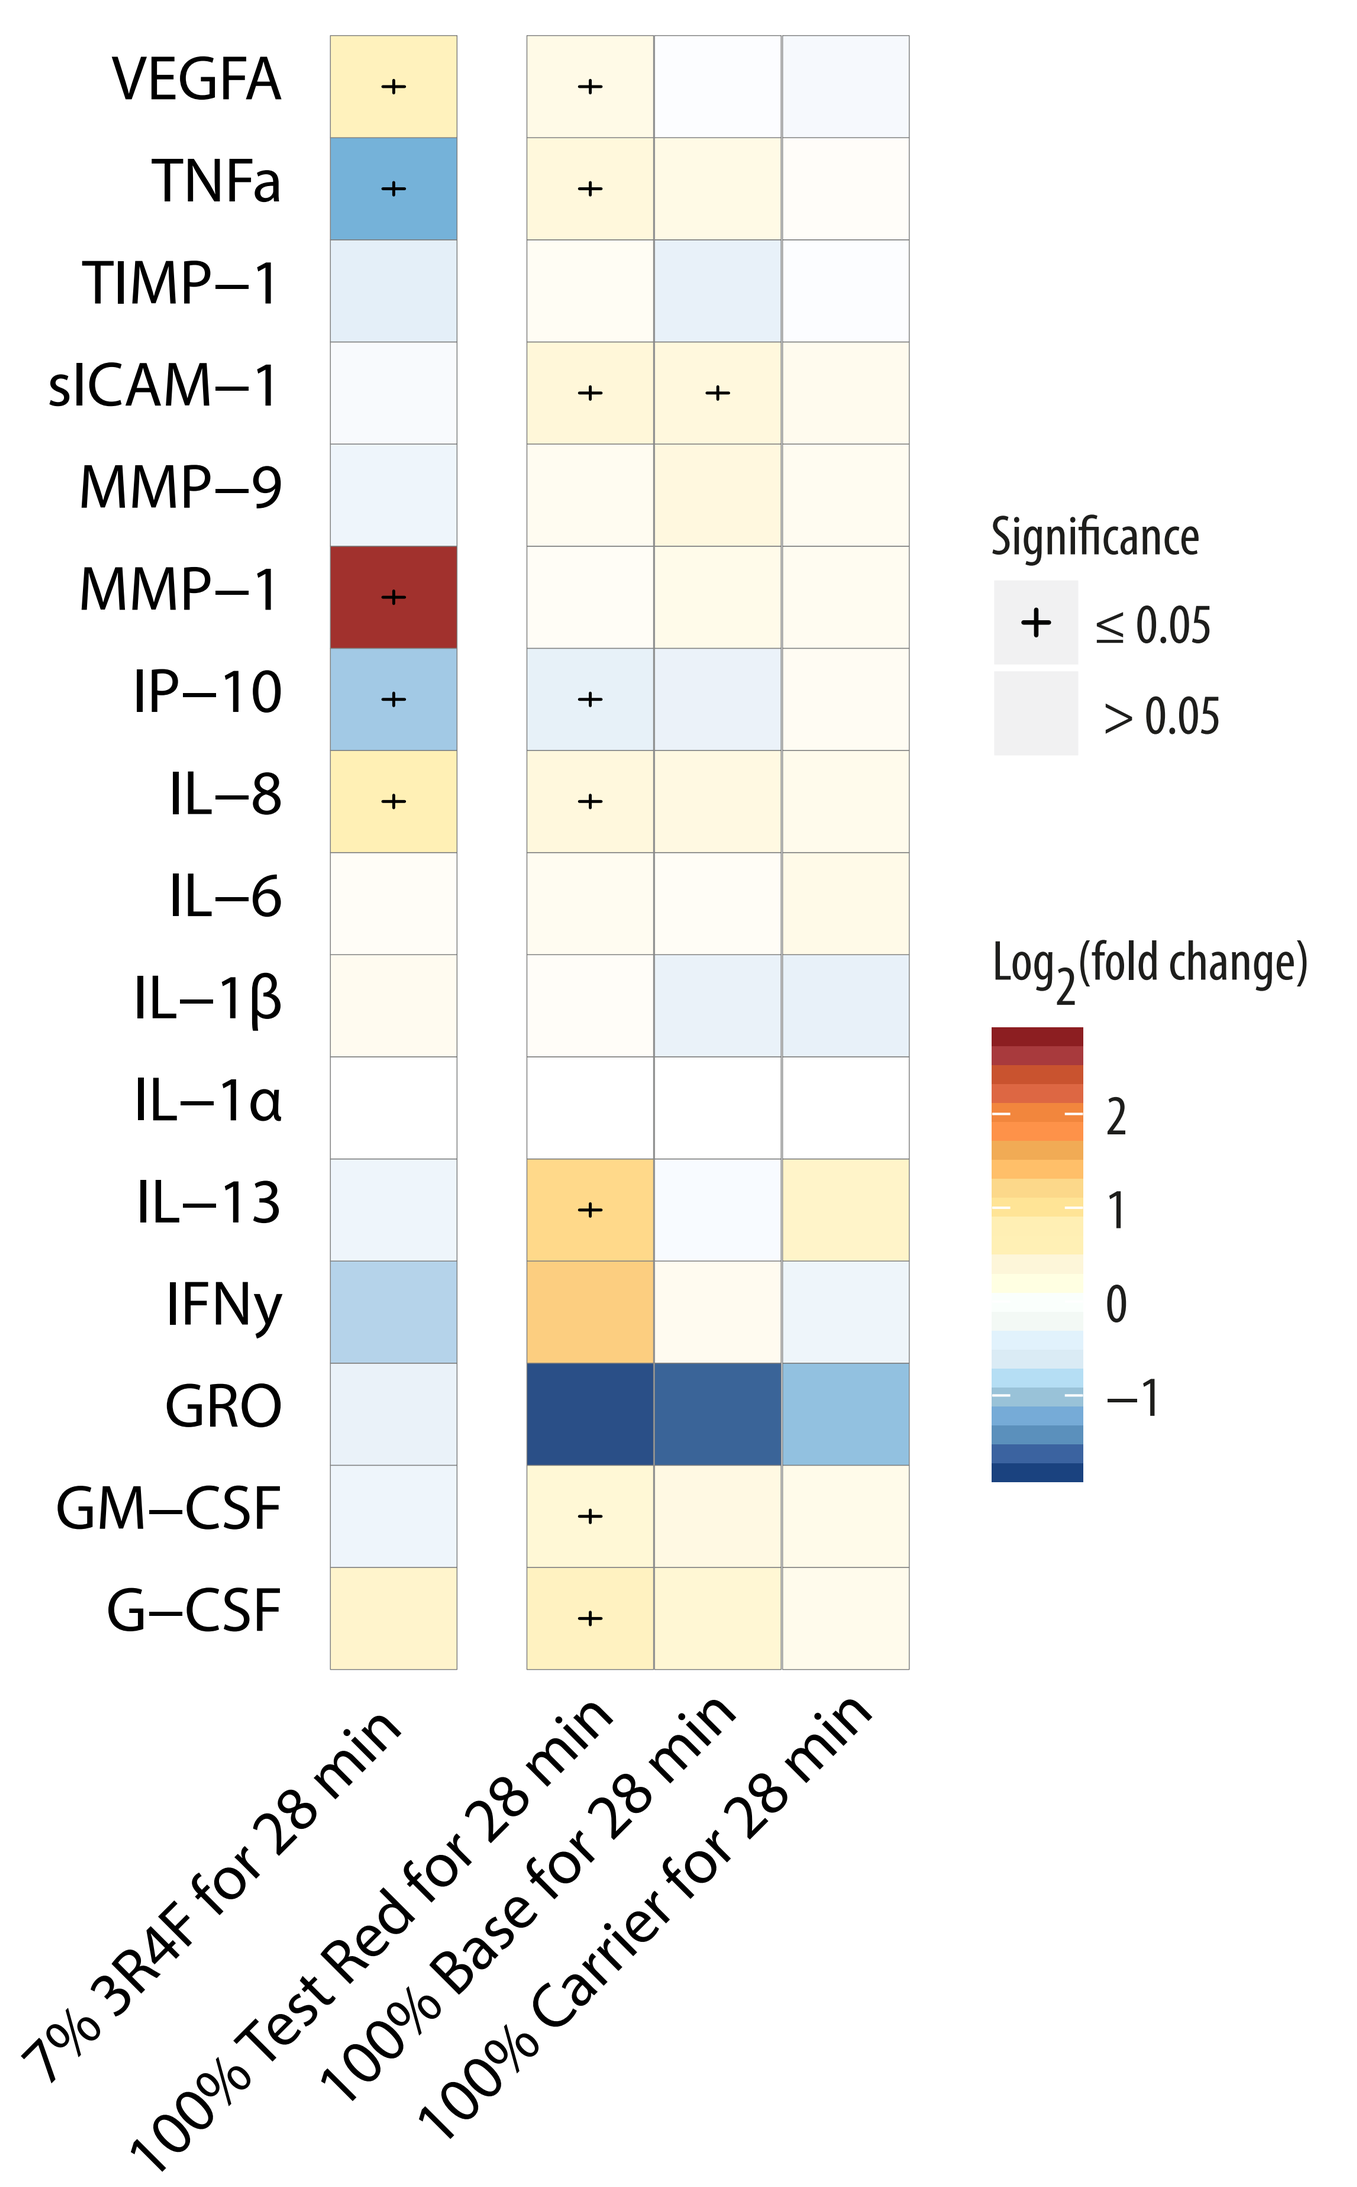
VEGFA
TNFa
TIMP−1
sICAM−1
MMP−9
MMP−1
IP−10
IL−8
IL−6
IL−1β
IL−1α
IL−13
IFNγ
GRO
GM−CSF
G−CSF
7% 3R4F for 28 min
100% Test Red for 28 min
100% Base for 28 min
100% Carrier for 28 min
Significance
+ ≤ 0.05
> 0.05
Log2(fold change)

Fold changes of the concentrations of secreted inflammatory mediators are shown. Exposure to 7% 3R4F CS for 28 minutes (112 puffs) markedly increased the concentrations of the mediators in the basolateral medium compared with the levels following exposure to air. Exposure to Test Red, Base, or Carrier aerosols generally elicited smaller changes in the mediator concentrations compared with the changes following exposure to 7% 3R4F CS for the same duration.

Conclusions

In conclusion, when compared to air exposure, exposure to Test Red, Base, or Carrier EC aerosols resulted in a lower impact than exposure to CS on in vitro human organotypic small airway epithelial cultures. Overall, marked differences in the measured endpoints (culture morphology, ciliary beating frequencies, secretion of inflammatory mediators, and global gene expression changes) were not observed following exposure to Test Red, Base, or Carrier EC aerosols. The observation further suggested minimal specific effects of nicotine or flavor ingredients in the alterations of various biological endpoints measured in the study.

References

BOUE, S., et al. 2015. Causal biological network database: a comprehensive platform of causal biological network models focused on the pulmonary and vascular systems. Database, 2015, bav030. **HOENG, J., et al. 2014.** Case study: the role of mechanistic network models in systems toxicology. Drug discovery today, 19, 183-192. **MARTIN, F., et al. 2014.** Quantification of biological network perturbations for mechanistic insight and diagnostics using two-layer causal models. BMC Bioinformatics, 15, 238. **MARTIN, F., et al. 2012.** Assessment of network perturbation amplitudes by applying high-throughput data to causal biological networks. BMC systems biology, 6, 54.

Competing Financial Interest –
The research described in this poster was sponsored by Philip Morris International. The authors are employees of Philip Morris International & Altria Client Services.
*Former Altria Client Services employee.

Society of Toxicology, Annual Meeting 2019, Baltimore, MD, USA | 10-14 March