A Multicenter, Multiregional Study on Biological and Functional Changes in Healthy Adult Smokers During One Year of Continuous Smoking Abstinence

PMI SCIENCE
PHILIP MORRIS INTERNATIONAL

L. Felber Medlin, C. T. Tran, W. T. Ng, N. Lama, B. Taranu, E. Pierri, P. Picavet, G. Baker, C. Haziza, F. Lüdicke

PMI R&D, Philip Morris Products S.A., Quai Jeanrenaud 5, CH-2000 Neuchâtel, Switzerland

Introduction and Objectives

The harm from smoking results mainly from long-term exposure to harmful and potentially harmful constituents (HPHC) in cigarette smoke generated by the combustion of tobacco. Smoking cessation (SC) is the most effective way to reduce the harm and risk of smoking-related diseases. In most SC studies, the main focus is on the rate of successful quitting for the SC approach/treatment tested; only limited information on multiple short- to long-term functional/biological changes following SC is available in the literature.

The overall study aim was to assess, over a one-year period of continuous smoking abstinence, the reversibility of the harm caused by smoking by assessing changes in clinical risk endpoints (CRE) linked to the pathophysiological pathways underlying the development of smoking-related diseases.

These CREs were selected according to epidemiological evidence that the endpoints are associated with smoking-related diseases and the expectation that these effects are reversed by SC over a period of time within the study duration.

Methods

Study Design

This was a multicenter, multiregional (U.S., Europe, Japan) SC study in healthy adult smokers planning to quit smoking within the next 30 days who were asked to continuously abstain from smoking during a one-year period in an ambulatory setting.

To support the subjects to stop smoking, SC support, including counseling and behavioral support, was provided throughout the study and upon subjects' request. Additionally, nicotine replacement therapy was provided at subjects' request and used as per country label for up to three months (+ two weeks).

Study Conduct

The study was approved by Independent Ethics Committees and Institutional Review Boards in all participating countries and was initiated in May 2015 and completed in May 2017. The study was conducted at 42 sites: 14 in the U.S., 13 in Europe, and 15 in Japan.

The study was conducted according to the International Conference on Harmonisation principles of Good Clinical Practice and registered on ClinicalTrials.gov (NCT02432729).

Main Eligibility Criteria

- Subjects understood the information provided for the study and signed the Informed Consent Form.
- Subjects smoked ≥10 commercially available cigarettes per day for the last 12 months and had been smoking for ≥ 10 years.
- Subjects were judged healthy by the Investigator.
- Subjects were 30 to 65 years old (inclusive).
- Female subjects were not pregnant or breastfeeding.
- Subjects were willing to quit smoking within the next 30 days.
- Subjects accepted continuous smoking abstinence for 52 weeks.

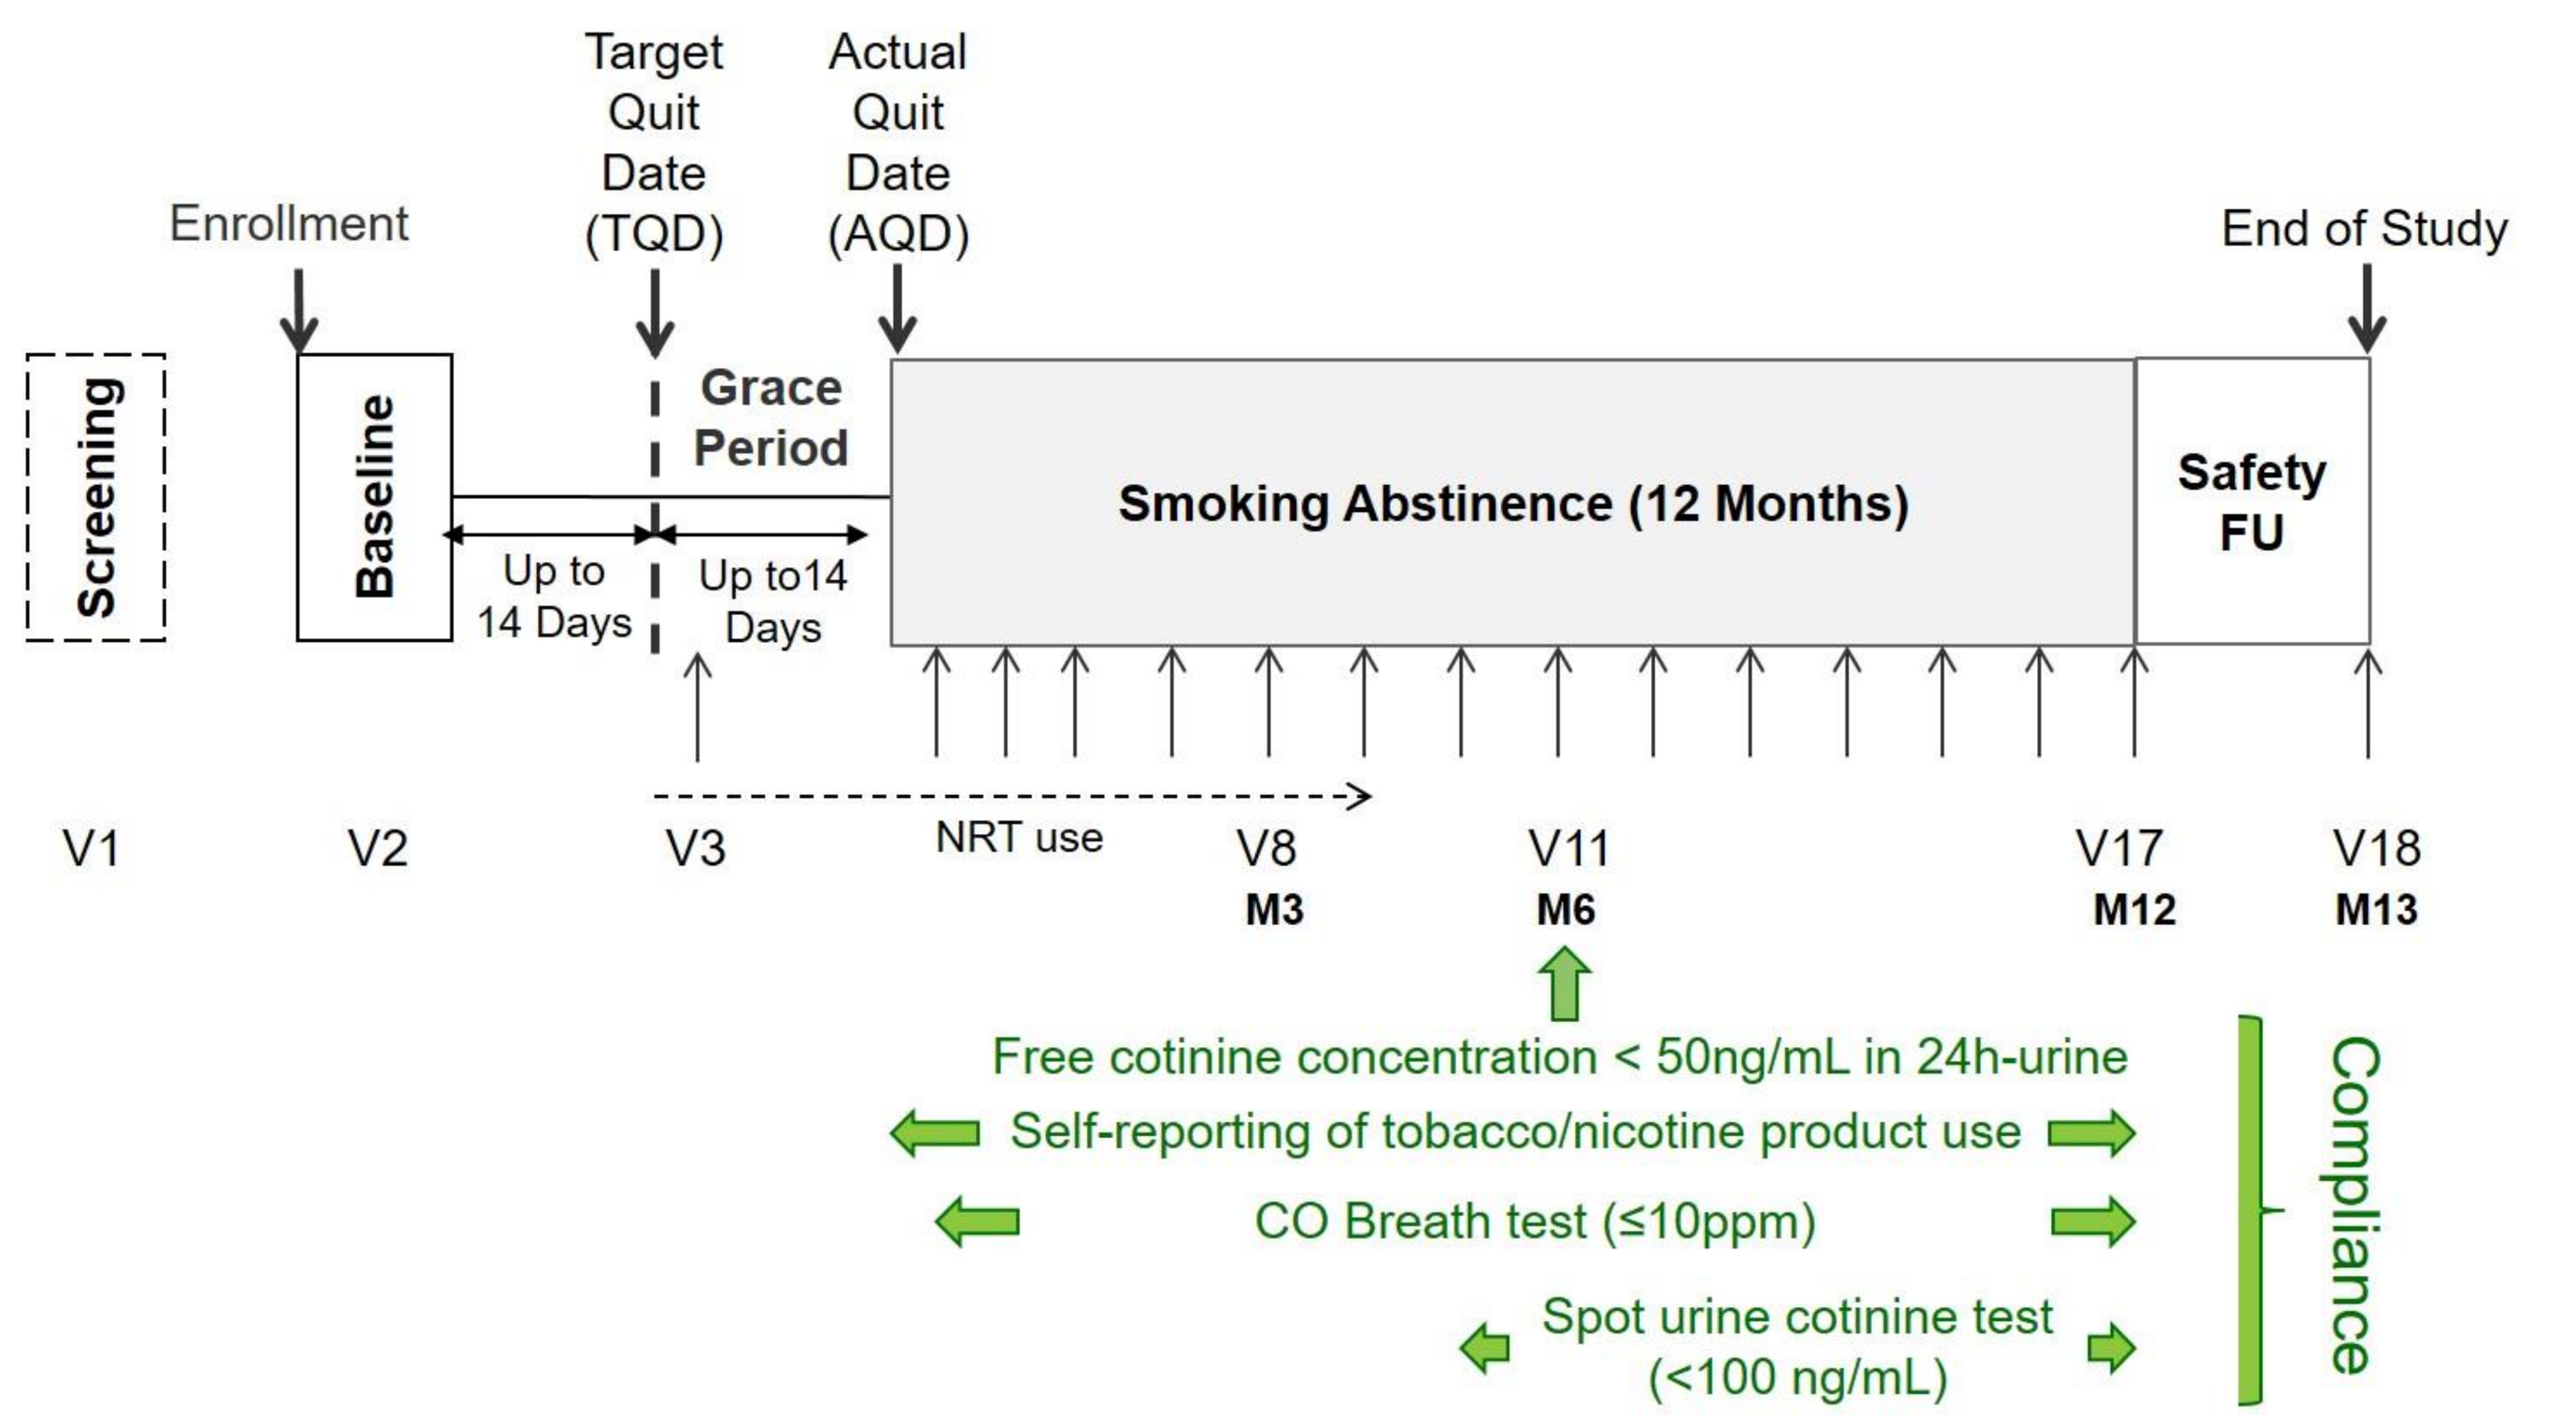
Enrollment
Target Quit Date (TQD)
Actual Quit Date (AQD)
End of Study
Screening
Baseline
Grace Period
Smoking Abstinence (12 Months)
Safety FU
Up to 14 Days
Up to14 Days
NRT use
V1
V2
V3
V8
M3
V11
M6
V17
M12
V18
M13
Free cotinine concentration < 50ng/mL in 24h-urine
Self-reporting of tobacco/nicotine product use
CO Breath test (≤10ppm)
Spot urine cotinine test (<100 ng/mL)
Compliance

Figure 1: Study Design.

Definitions

Change from Baseline, where Baseline value is defined as the last available value prior to both TQD and AQD.
% change from Baseline, where Baseline value is defined as the last available value prior to both TQD and AQD.
RC = Relative change from Baseline, as estimated from the ratio of the geometric means.

Results

Subject Disposition

Of the 1,184 enrolled subjects, 358 (30.2%) were continuously abstinent from smoking from AQD to Month 12.

Table 1: Study Populations.

Number of Subjects	Global	US	EUR	JP
Planned	950			
Screened	2089			
Enrolled	1184			
Quitters*	811	178	387	246
Continuously abstinent at Month 3	720			
Continuously abstinent at Month 6	450			
Continuously abstinent at Month 12	358	73	170	115

*Quitters = All enrolled subjects with at least one valid non-safety assessment after AQD and with no major protocol deviations impacting the overall subject evaluability.

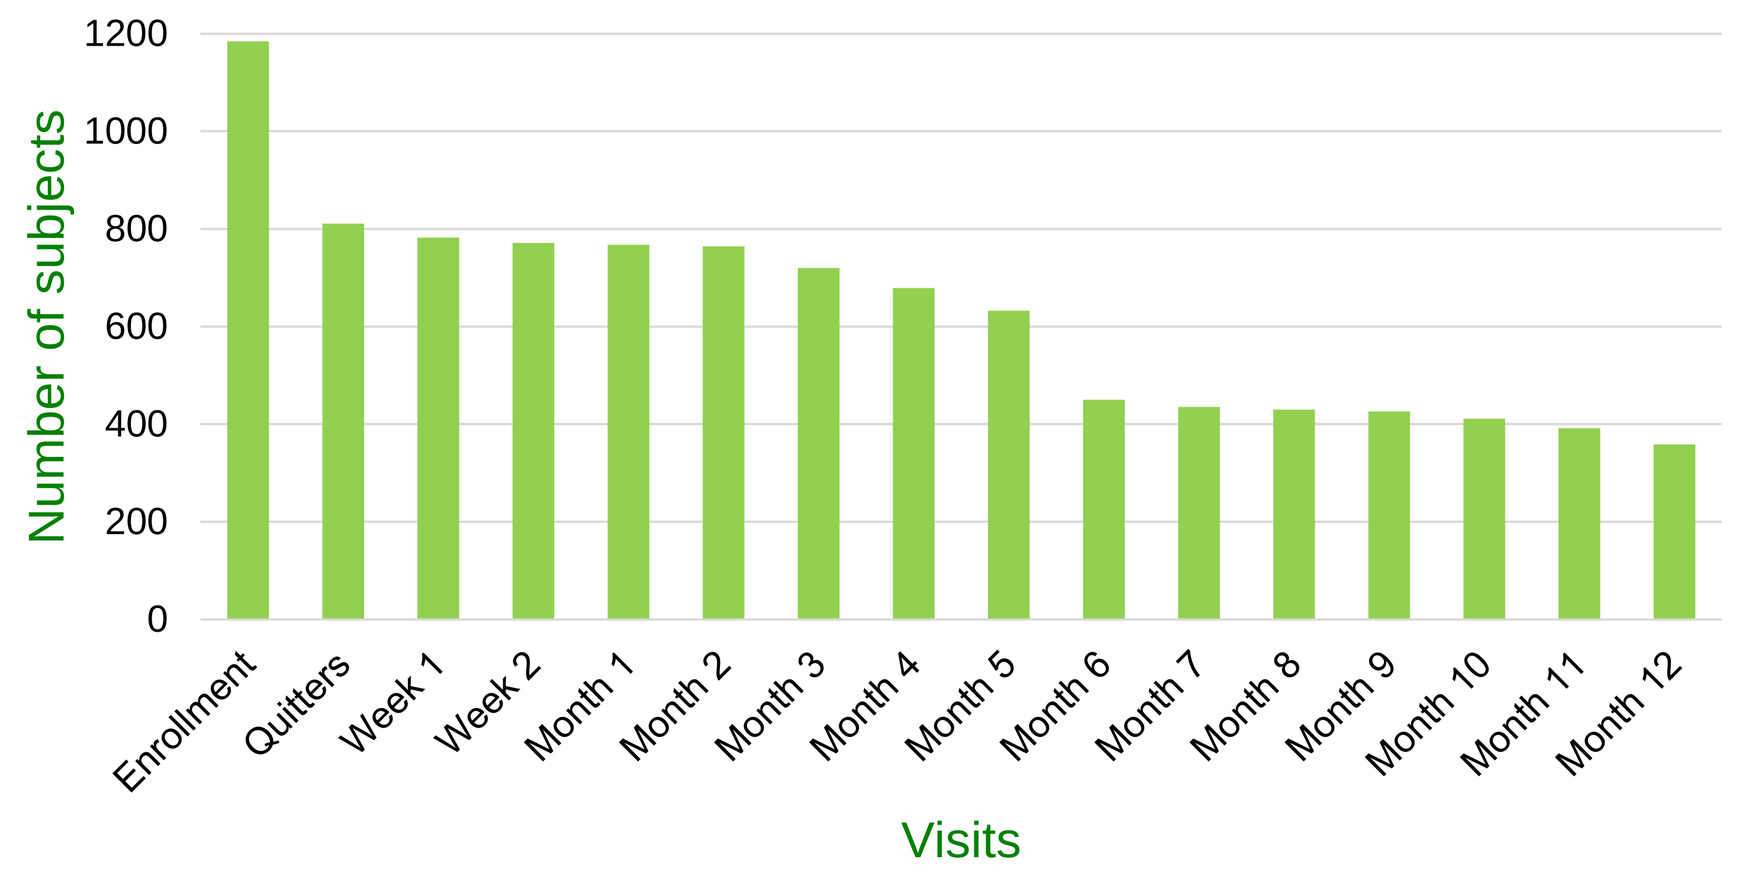
Number of subjects
Visits
Enrollment
Quitters
Week 1
Week 2
Month 1
Month 2
Month 3
Month 4
Month 5
Month 6
Month 7
Month 8
Month 9
Month 10
Month 11
Month 12

Figure 2: Number of Subjects Continuously Abstinent.

Demographics

Table 2: Baseline Characteristics.

Characteristics	Enrolled Population (N=1184)	Abstinent Subjects at Month 12 (N=358)			
		Overall	US	EUR	JP
Male: n (%)	593 (50.1)	181 (50.6)	24 (32.9)	84 (49.4)	73 (63.5)
Female: n (%)	591 (49.9)	177 (49.4)	49 (67.1)	86 (50.6)	42 (36.5)
Age (years): Mean (SD)	44.1 (8.87)	43.8 (9.21)	45.1 (8.54)	42.6 (9.26)	44.7 (9.41)
Caucasian: n (%)	678 (57.3)	203 (56.7)	37 (50.7)	166 (97.6)	0
Not Caucasian: n (%)	505 (42.7)	155 (43.3)	36 (49.3)	4 (2.4)	115 (100)
BMI (kg/m2): Mean (SD)	25.4 (4.02)	24.8 (3.75)	26.9 (3.54)	25.2 (3.71)	23 (3.06)
Smoking duration (years) : Mean (SD)	23.7 (8.61)	22.8 (8.76)	24.2 (8.42)	22 (8.71)	23.2 (8.98)
Smoking intensity over the past year (cig/day): Mean (SD)	17.9 (6.65)	16.6 (5.27)	17.2 (5.59)	16.6 (5.2)	16.2 (5.16)

Biomarkers of Exposure (BoExp) to Various HPHCs

Levels of all BoExp to HPHCs were substantially reduced, by 54.5% to 97.9%.

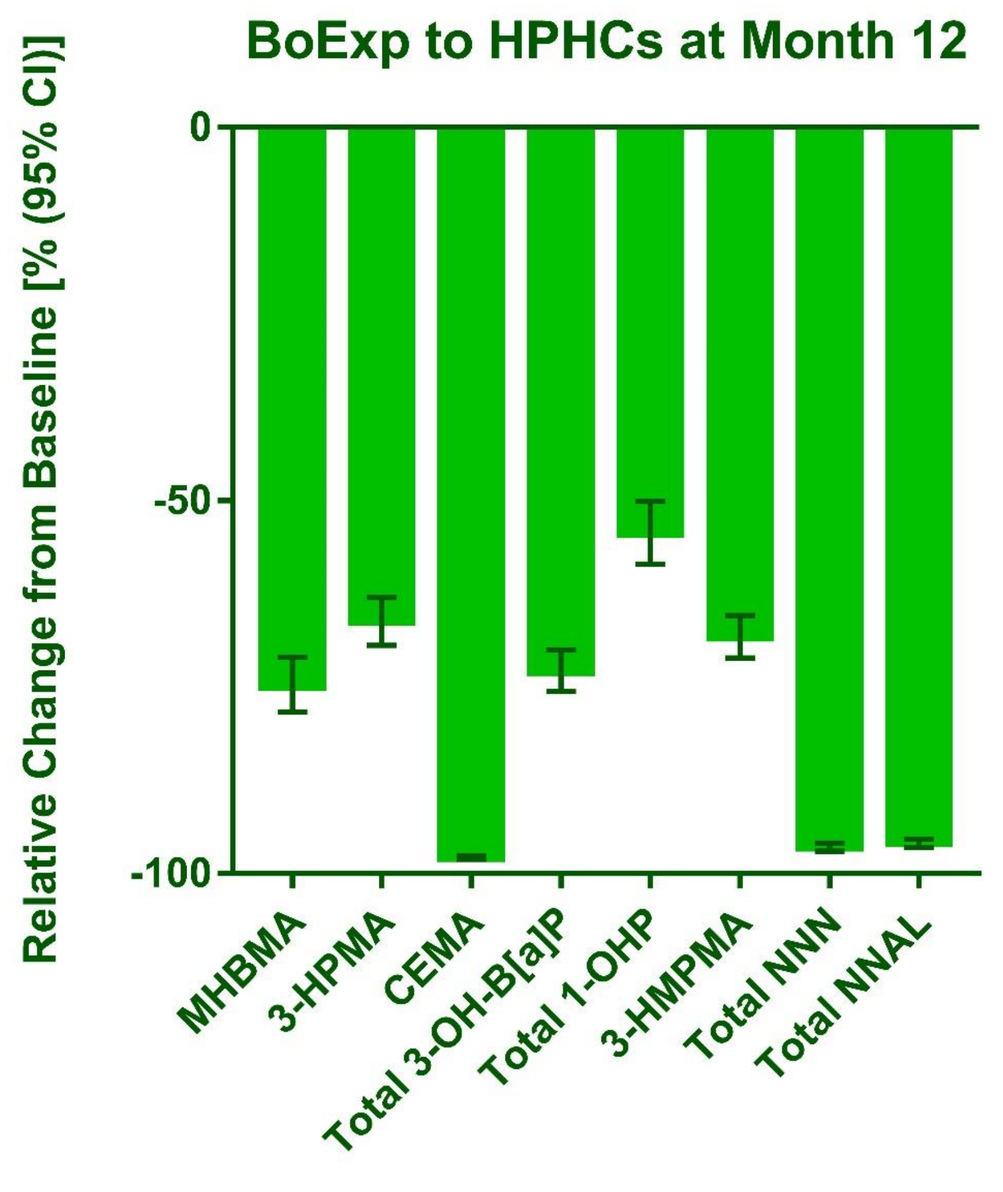
BoExp to HPHCs at Month 12
Relative Change from Baseline [% (95% CI)]
MHBMA
3-HPMA
CEMA
Total 3-OH-B[a]P
Total 1-OHP
3-HMPMA
Total NNN
Total NNAL

Abbreviation	Biomarker	HPHCs
MHBMA	Monohydroxybutenylmercapturic acid	1,3-Butadiene
3-HPMA	3-Hydroxypropylmercapturic acid	Acrolein
CEMA	2-Cyanoethylmercapturic acid	Acrylonitrile
Total 3-OH-B[a]P	Total 3-hydroxylbenzo(a)pyrene	Benzo[a]pyrene
Total 1-OHP	Total 1-Hydroxypyrene	Pyrene
3-HMPMA	3-Hydroxy-1-methylpropylmercapturic acid	Crotonaldehyde
Total NNN	Total N-nitrosonornicotine	N-nitrosonornicotine
Total NNAL	Total 4-(methylnitrosamino)-1-(3-pyridyl)-1-butanol	4-(methylnitrosamino)-1-(3-pyridyl)-1-butanone

Figure 3: BoExp to HPHCs.

CREs Associated with Cardiovascular Diseases (CVD)

4A

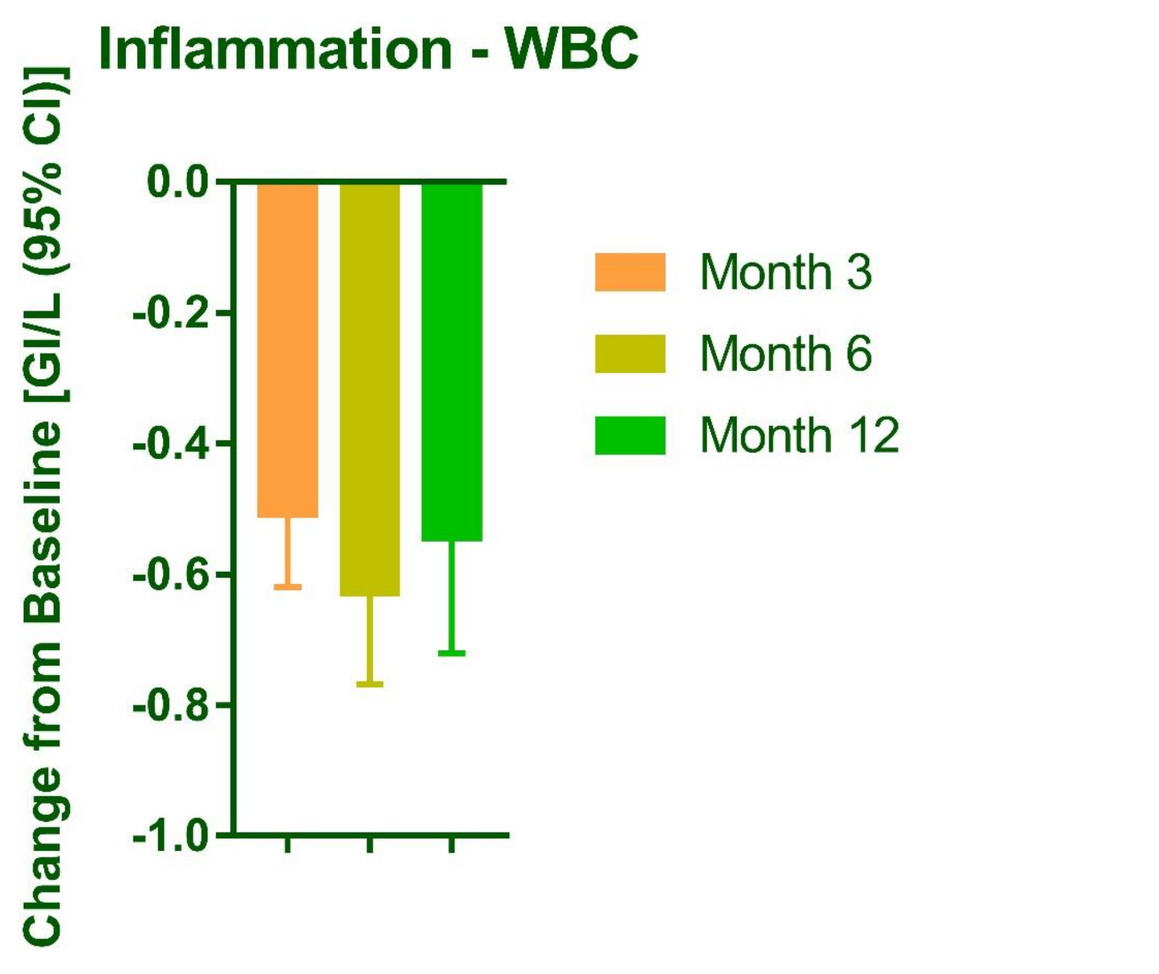
Inflammation - WBC
Change from Baseline [GI/L (95% CI)]
Month 3
Month 6
Month 12

4B

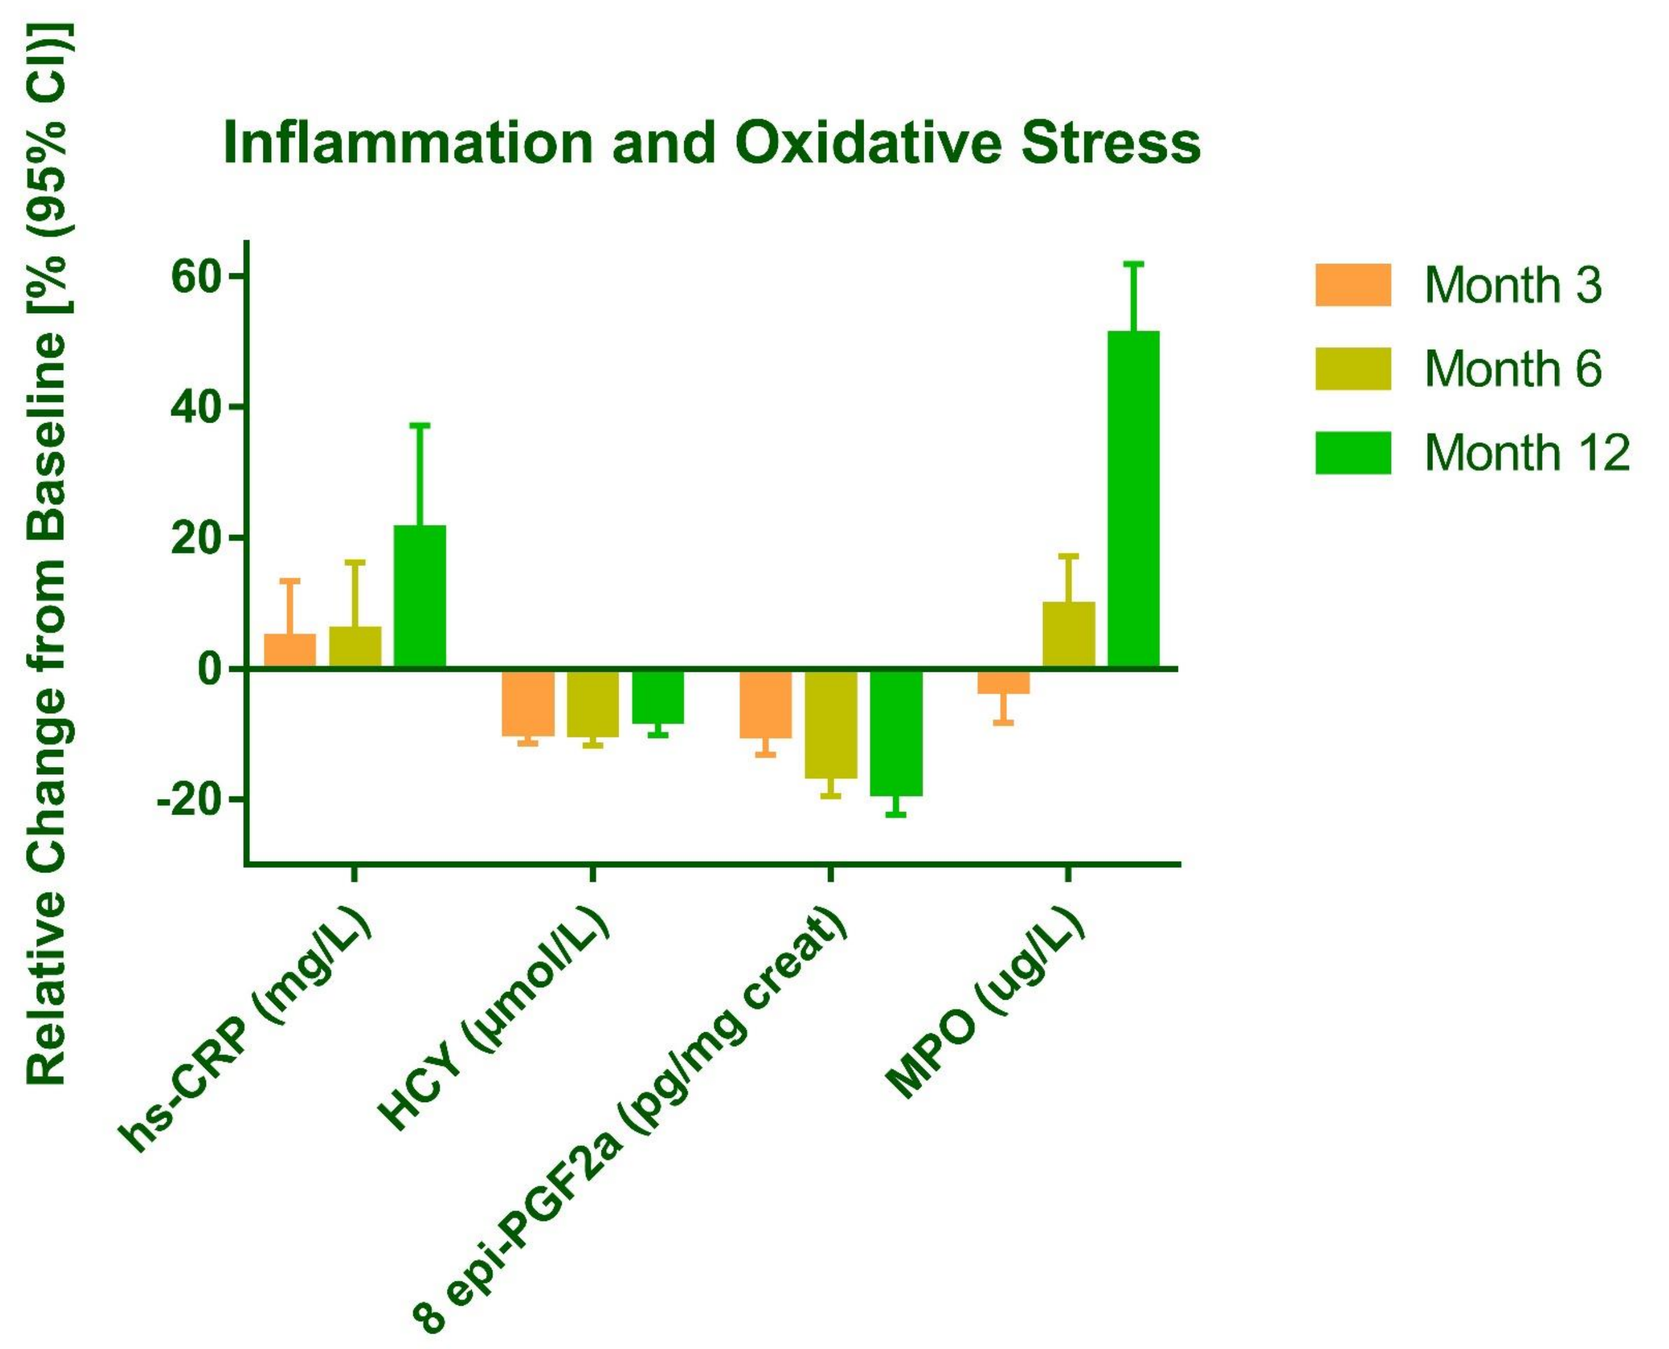
Inflammation and Oxidative Stress
Relative Change from Baseline [% (95% CI)]
hs-CRP (mg/L)
HCY (umol/L)
8 epi-PGF2a (pg/mg creat)
MPO (ug/L)
Month 3
Month 6
Month 12

Figure 4A and 4B: Cardiovascular CRE.
Abbreviations. White Blood Cell (WBC); High Sensitivity C-Reactive Protein (hsCRP); Homocysteine (HCY); 8-epi-prostaglandin-alpha (8-epi-PGF2a); Myeloperoxidase (MPO).

Results

CREs Associated with CVDs

5A

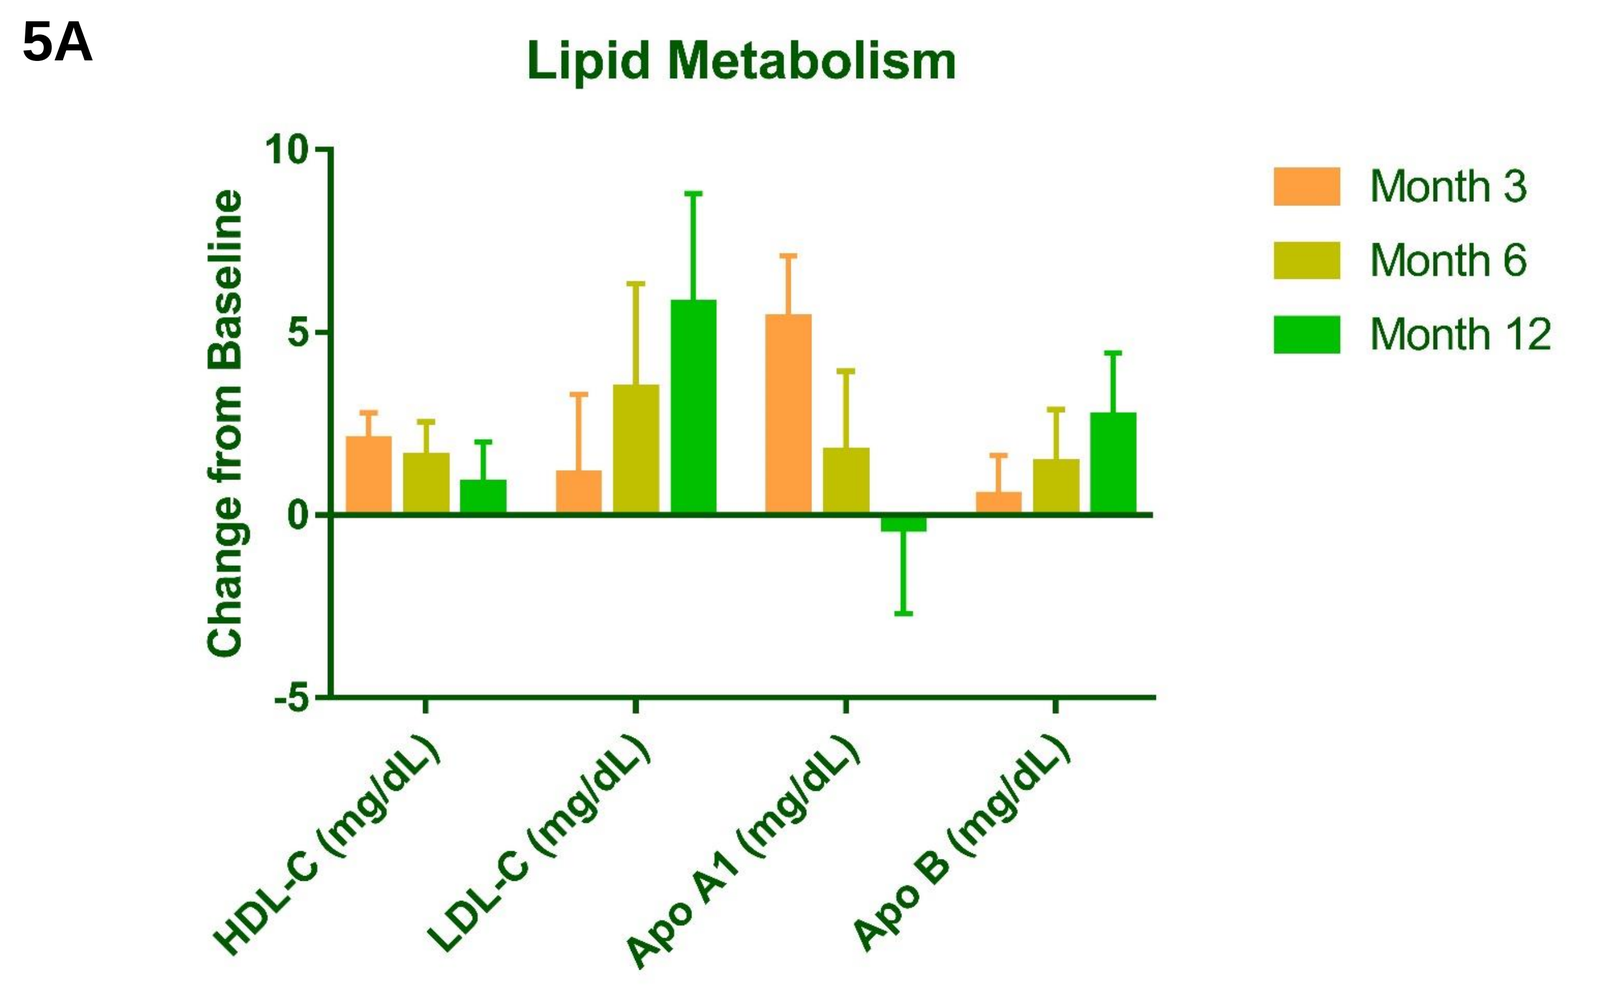
Lipid Metabolism
Change from Baseline
HDL-C (mg/dL)
LDL-C (mg/dL)
Apo A1 (mg/dL)
Apo B (mg/dL)
Month 3
Month 6
Month 12

5B

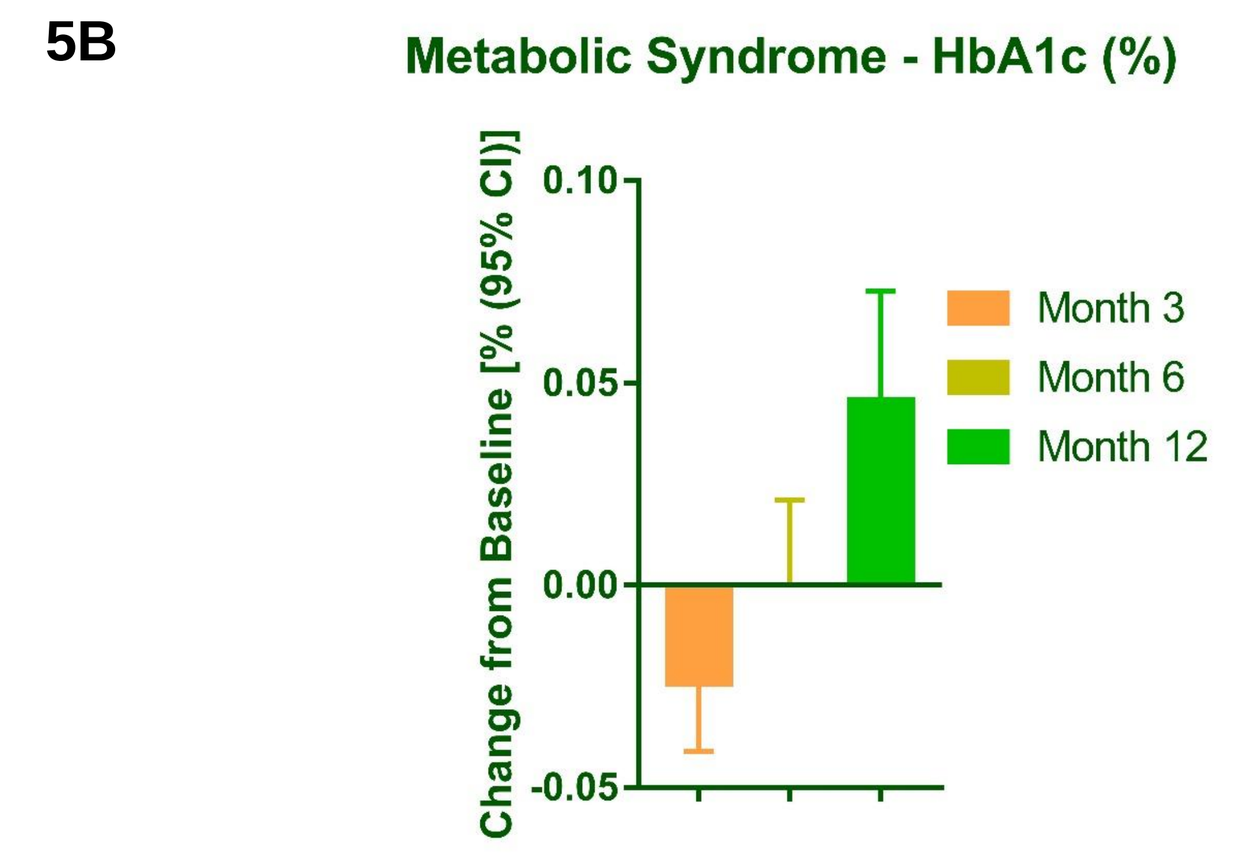
Metabolic Syndrome - HbA1c (%)
Change from Baseline [% (95% CI)]
Month 3
Month 6
Month 12

5C

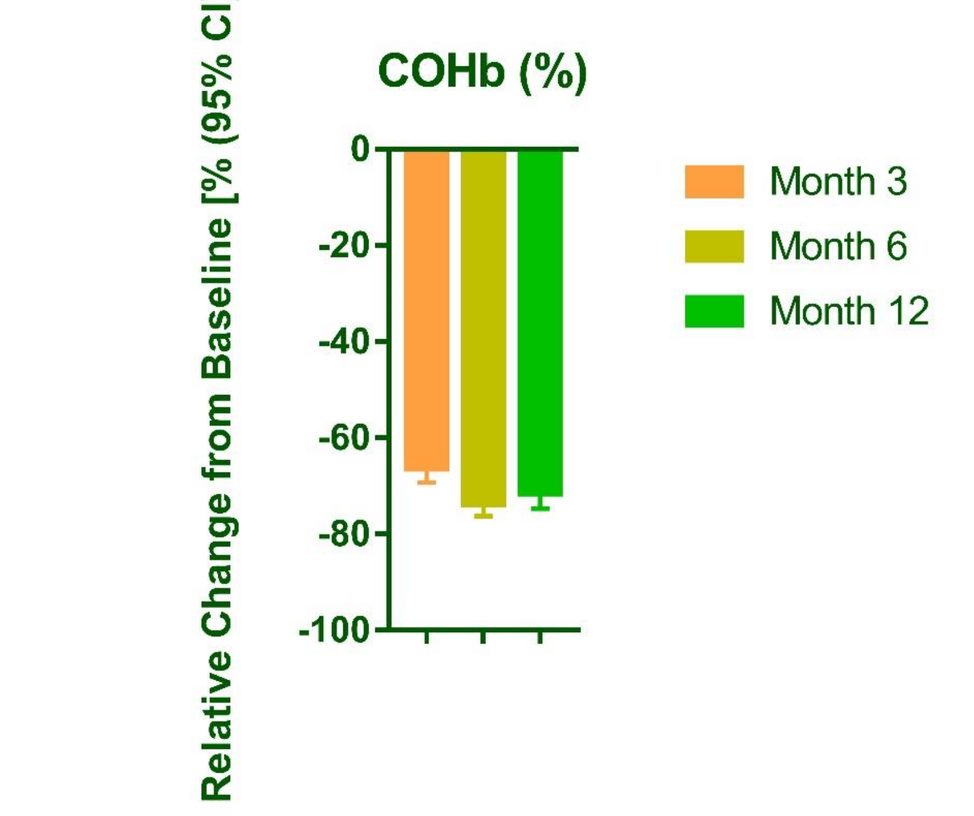
COHb (%)
Relative Change from Baseline [% (95% CI)]
Month 3
Month 6
Month 12

5D

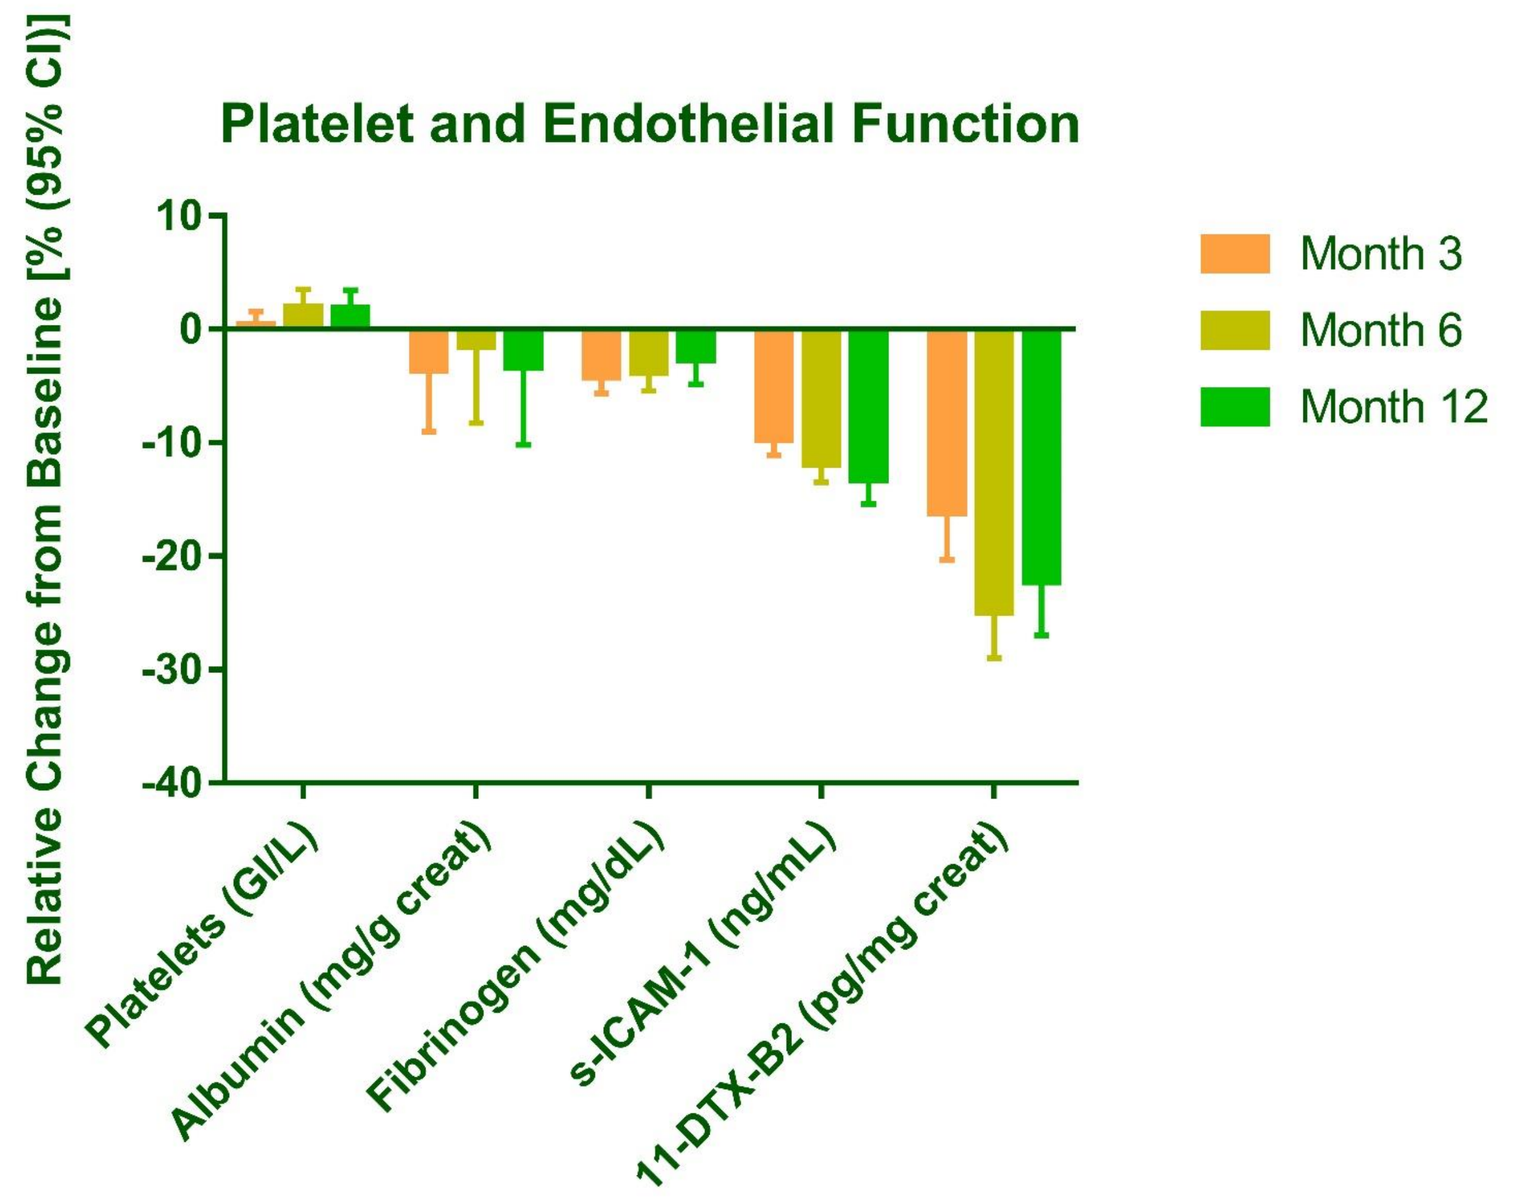
Platelet and Endothelial Function
Relative Change from Baseline [% (95% CI)]
Platelets (GI/L)
Albumin (mg/g creat)
Fibrinogen (mg/dL)
s-ICAM-1 (ng/mL)
11-DTX-B2 (pg/mg creat)
Month 3
Month 6
Month 12

Figure 5 A–D: Cardiovascular CRE.
Abbreviations. High and low density lipoprotein cholesterol (HDL-C, LDL-C); Apolipoprotein A1 and B (Apo A1 and Apo B); glycosylated hemoglobin (HbA1c); carboxyhemoglobin (COHb); soluble intercellular adhesion molecule-1 (sICAM-1); 11-dehydrothromboxane B2 (11-DTX-B2).

CREs Associated with Respiratory Diseases

Most of the lung function parameters assessed pre- and post-bronchodilator remained similar to Baseline over the course of the study, except a slight decrease in TLC, FRC, and RV.

Table 3: Cough (VAS Questionnaire)

Number of subjects who experienced regular need to cough - n(%)	
Baseline	242 (33.6)
Month 3	109 (15.1)
Month 6	45 (10.0)
Month 12	40 (11.2)

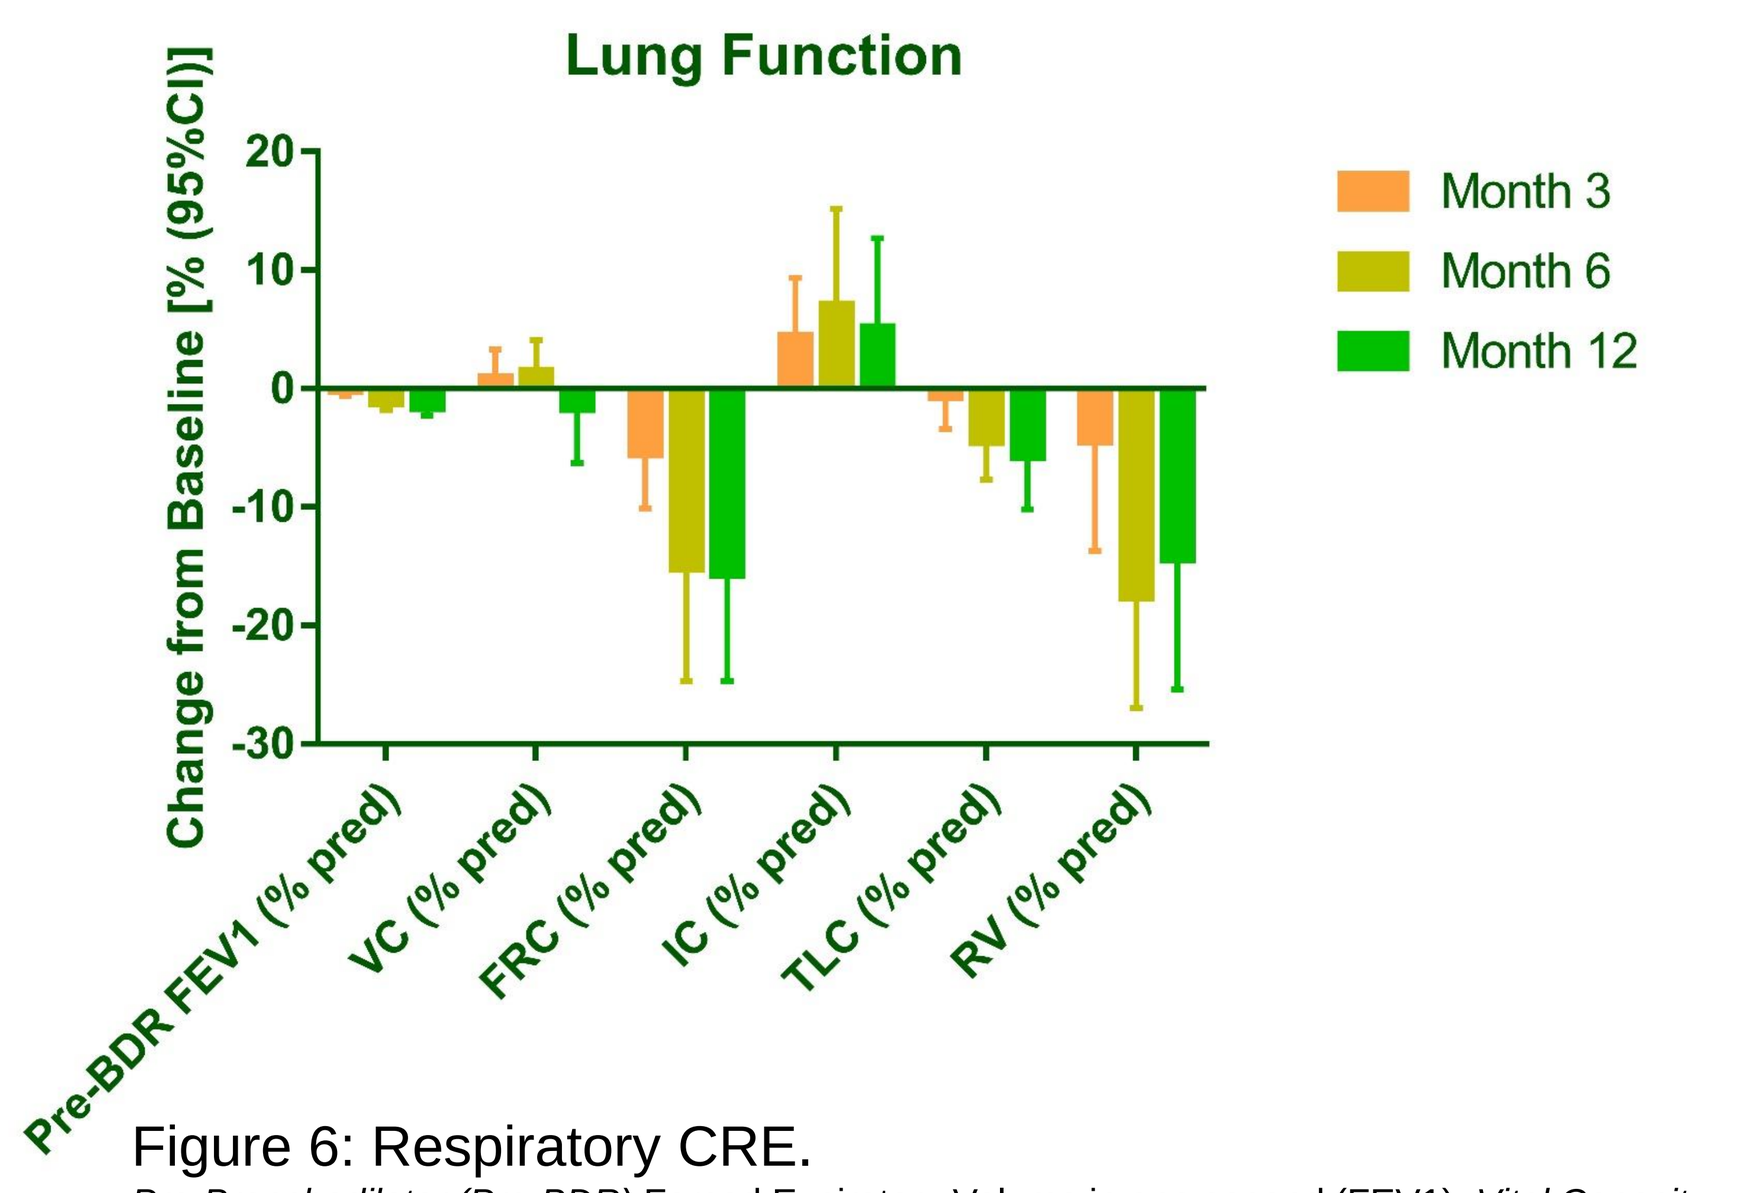
Lung Function
Change from Baseline [% (95%CI)]
Pre-BDR FEV1 (% pred)
VC (% pred)
FRC (% pred)
IC (% pred)
TLC (% pred)
RV (% pred)
Month 3
Month 6
Month 12

Figure 6: Respiratory CRE.
Pre-Bronchodilator (Pre-BDR) Forced Expiratory Volume in one second (FEV1); *Vital Capacity (VC); Functional Residual Capacity (FRC); Inspiratory Capacity (IC); Total Lung Capacity (TLC); Residual Volume (RV).*

Safety

Overall, 20 serious adverse effects (SAE) were reported by 17 subjects (1.4%), with one fatal SAE reported. No SAEs were related to study procedures. The majority of adverse effects (AE) were mild or moderate in severity. Overall in the study, there was an increase from Baseline at Month 3, Month 6, and Month 12 in both mean body weight (2.24 kg, 3.46 kg, and 4.06 kg, respectively) and BMI (0.776 kg/m^2, 1.18 kg/m^2, and 1.41 kg/m^2, respectively).

Conclusions

- After continuous smoking abstinence over 12 months, most CREs associated with CVDs, respiratory diseases, and carcinogenicity showed favorable changes.
- Substantial decreases (ranging from –54.5% to –97.9% from baseline) in the BoExp levels to HPHCs were observed after 12 months of continuous SC.
- Most AEs were assessed as mild to moderate in severity.
- The continuous abstinent rate among the 1,184 enrolled subjects was 30% (= 358 subjects who successfully quit smoking for one year).

These results indicate that continuous abstinence from smoking for one year leads to a substantial reduction in exposure to HPHCs and favorable changes in CREs involved in multiple mechanistic pathways and biological functions, such as lipid metabolism, inflammation, or oxidative stress, that are likely to contribute to the reduction of risk of developing smoking-related diseases.

SRNT 25th Annual Meeting, San Francisco, CA, USA, February 20–23, 2019

Competing Financial Interest – The research described in this poster was sponsored by Philip Morris International.