Exploring Discriminant Capacity of Urinary CEMA as Combustion Marker in Tobacco Users - A Population Pharmacokinetic Approach

A. Claussen[1], A. Donelli[2], W. Copalu[1], N. Lama[2], R. Weitkunat[2], C. Haziza[2], F. Lüdicke[2]

[1] Certara Strategic Consulting, Rue du Colonel Moll 3, Paris 75017, France

[2] PMI R&D, Philip Morris Products S.A., Quai Jeanrenaud 5, CH-2000 Neuchâtel, Switzerland

Introduction and Objectives

Acrylonitrile is included in the list established by the Food and Drug Administration on harmful and potentially harmful constituents (HPHC) in tobacco products and tobacco smoke, and is classified as possibly carcinogenic to humans by the International Agency for Research on Cancer. Acrylonitrile is generated at temperatures ranging from 500°C to 800°C [1], making the 2-cyanoethylmercapturic acid (CEMA) metabolite, a biomarker of exposure to acrylonitrile, a good candidate as biomarker of verification for subject reported product use (e.g. in clinical studies) and, in particular for a diagnostic marker to distinguish cigarettes (CC) smoking from non-combustible tobacco product use, such as the Tobacco Heating System (THS), a heat-not-burn tobacco product that heats tobacco to temperatures up to 350°C, well below the temperature required for combustion. In order to minimize the variability of CEMA in urine, urinary creatinine concentration is most commonly used to normalize the analyte concentration ($CEMA_{creat}$).

The objective of this analysis was to assess the ability of 24-hour and spot urine $CEMA_{creat}$ to discriminate CC use and smoking abstinence (SA) status, and CC from THS use.

To this aim, a population pharmacokinetics (PK) analysis was performed to describe acrylonitrile absorption following CC smoking or use of THS, its metabolization into CEMA, and its final elimination. Discrimination performance of $CEMA_{creat}$ was evaluated in data simulated from a population PK model.

Methods

Dataset

Data were pooled for analysis from four exposure reduction studies [3], conducted as randomized, three-arm parallel control group design studies assessing exposure to HPHCs following THS use compared with CC use, using SA as benchmark for five days in a confined setting.

Urine samples for the determination of CEMA and creatinine concentrations in urine were analyzed using a validated liquid chromatography coupled to tandem mass spectrometry assay method [2].

Data were split 50-50 into learning and validation sets. The learning set was used for model building, covariate exploration, and internal model qualification. The validation dataset was used for external model validation.

Population PK Model

The base model for $CEMA_{creat}$ was two-compartment linear disposition with first order absorption and elimination, and log additive residual error. Product use events were individually modelled as boluses during Day –1 through Day 5. CC exposure before t_0 at Day –1 was modeled as infusion during the previous 30 days.

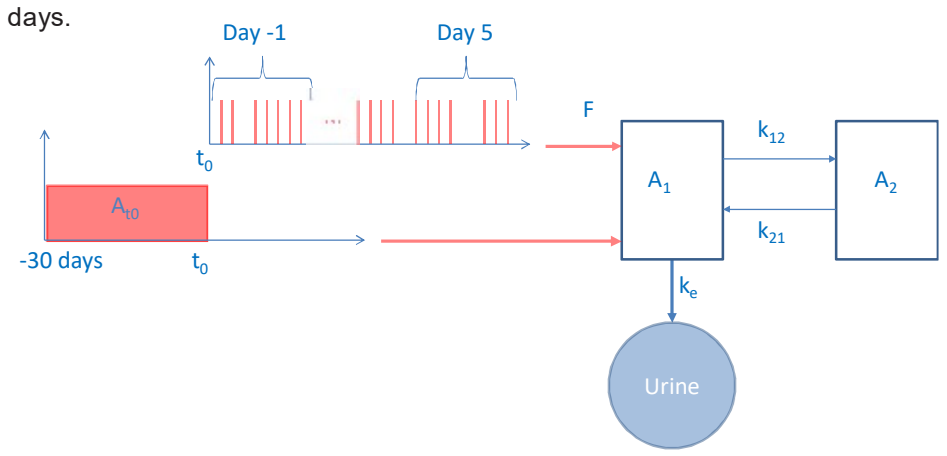
Day -1
Day 5
F
t_0
A_{t0}
-30 days
t_0
A_1
A_2
k_{12}
k_{21}
k_e
Urine

Figure 1: Base model parameters.

- A_1, A_2: model compartments
- k_{12}, k_{21}: inter-compartmental rate constants
- k_e: terminal elimination rate constant
- F: bioavailability of THS relative to CC
- t_0: start time of product use events recorded at study Day –1
- A_{t0}: bioavailability of pre-t_0 exposure

Graphical exploratory variable analyses were used to identify possible sources of variability of PK parameters. Modeling and simulations were conducted using the non-linear mixed-effect method. Goodness-of-fit (GOF) diagnostics and posterior visual predictive checks (VPC) were used to evaluate the model adequacy.

Sub-model	Equations
Product use	$dA_1/dt = -Ke\ A_1 - k_{12} A_1 + k_{21} A_2$ $dA_2/dt = k_{12}A_1 - k_{21}A_2$ with $k_{21} = k_{21}(tv) \times \Delta K_{21}$ (CC vs. non-CC) $dA_0/dt = Ke\ A_1$ with $ke = Ke(tv) \times \Delta Ke(CEMA_{crB}) \times \Delta Ke$ (US vs. non-US)
Background pre-t_0 Exposure	30 days x A_{to}
Total	Total $CEMA_{creat}$ = pre-t_0 exposure + product use

Table 1: Population PK model equations.

Discrimination Performance Evaluation Methods

The potential of $CEMA_{creat}$ as a diagnostic marker to distinguish between smoking and non-combustible tobacco use was explored by means of receiver operating characteristics (ROC) analysis.

$CEMA_{creat}$ cut-offs were selected to minimize the overall misclassification probability. Additional thresholds were derived to minimize the false positive error, because the consequences of wrongly missing subjects using THS may be more relevant in certain applications.

Results

The PK population consisted of 632 subjects. No subject from the pooled set was excluded from the analysis.

Variable		Unit	Learning Set (N=322)	Validation Set (N=310)
Sex - female		n (%)	149 (46.3%)	142 (45.8%)
Age		year	36.7 ± 10.7	36.7 ± 11
Weight		kg	70.2 ± 13.7	69.4 ± 13.9
ALT		IU/L	17.6 ± 8.2	18.1 ± 9.6
AST		IU/L	18.2 ± 4.7	18.4 ± 5.2
Total bilirubin		mg/dL	0.6 ± 0.2	0.6 ± 0.3
Creatinine CL (baseline)		mL/min	126.9 ± 32.8	122.8 ± 28.3
$CEMA_{BCR}$		ng/mg creat	99.4 ± 52.8	108.8 ± 58.7
Region	US	n (%)	79 (24.5%)	75 (24.2%)
	EU		82 (25.5%)	78 (25.2%)
	JP		161 (50%)	157 (50.6%)
Exposure	THS	n (%)	160 (49.7%)	155 (50.0%)
	CC		84 (26.1%)	79 (25.5%)
	SA		78 (24.2%)	76 (24.5%)

Table 2: Demographics of analysis sets.

Following graphical exploratory data analysis of covariates, the final population PK model included effects of baseline $CEMA_{creat}$ value ($CEMA_{crB}$) on the elimination rate (K_e), product type (CC vs. non CC) on the inter-compartmental rate from compartment 2 to 1 (K_{21}), and being from the U.S. region on elimination rate (K_e). Initial half-life was 2.1 hours (95% prediction interval (PI): 1.8, 2.3), and the terminal half-life was 212 hours (95% PI: 157.8, 266.5) for a typical subject not from the U.S. and median $CEMA_{crB}$ of 92 ng/mg creat.

Parameter	Estimate	95% PI	Effect Equation
A_{t0}	0.092	0.079, 0.105	
K_{12}	$0.209 h^{-1}$	$0.177, 0.241\ h^{-1}$	
K_{21}(tv)	$0.00925 h^{-1}$	$0.00818, 0.0103\ h^{-1}$	
K_e (tv)	$0.117 h^{-1}$	$0.0943, 0.141\ h^{-1}$	
F_{CC}	18.222	15.577, 20.868	
PerTHS	–4.385	–5.340, –3.430	$F_{THS}=F_{CC}\exp(Per_{THS}) = 0.23$
ΔK_e ($CEMA_{crB}$)	1.029	1.013, 1.045	$1+\Delta K_e\ (CEMA_{crB}/92^{(*)}-1)$
ΔK_e (US vs. non-US)	–0.61	–0.689, –0.531	$1+\Delta K_e$ (=0.39 for US)
ΔK_{21} (CC vs. non-CC)	–0.863	–0.947, –0.78	$1+\Delta K_{21}$ (=0.137 for CC)

Table 3: Estimates of population PK model parameters.
* The median $CEMA_{crB}$

The elimination rate (K_e) increased for higher $CEMA_{crB}$ (31% for a $CEMA_{crB}$ at 120 ng/mg), which would result in a decrease in AUC and a shorter half-life. K_e was also 61% lower for subjects in the U.S. K_{21} was 86% lower in CC users. The absorbed "dose" of one product consumption (bioavailability) was 18.22 for CC users and 0.23 for THS users. Thus, relative to CC, 1.3% acrylonitrile was absorbed by a THS user.

The GOF plots and individual fits showed good agreement between predicted and observed values, with no apparent bias. Based on the validation set, VPC revealed that the model adequately captured the median PK profile, especially at later time points (Days 4 and 5) where the subsequent biomarker qualification is focused. The population PK model characterized the kinetics of CEMA adequately.

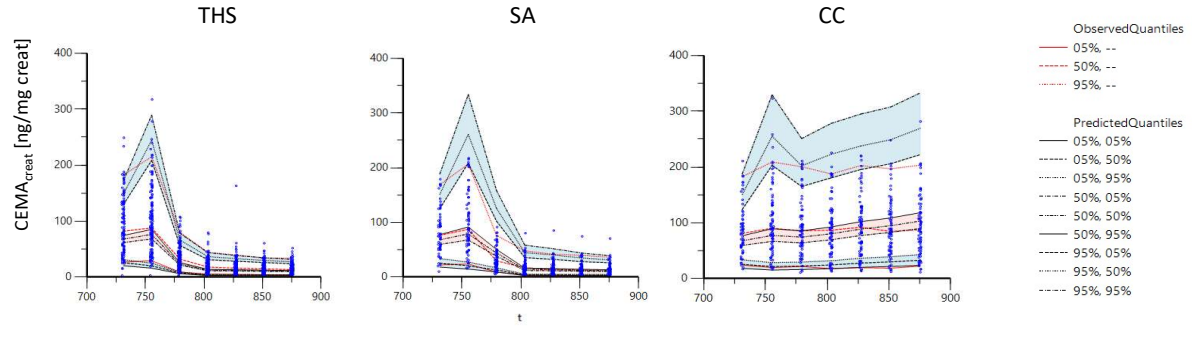
THS
SA
CC
$CEMA_{creat}$ [ng/mg creat]
t
ObservedQuantiles
05%, --
50%, --
95%, --
PredictedQuantiles
05%, 05%
05%, 50%
05%, 95%
50%, 05%
50%, 50%
50%, 95%
95%, 05%
95%, 50%
95%, 95%

Figure 2: Visual predictive checks on validation set.

Biochemical Verification of Exposure Using CEMA – Discrimination Performance Results

Qualification of $CEMA_{creat}$ as biomarker of verification to distinguish SA vs. CC and THS vs. CC was conducted on both original data (n=154, 163, 325 for SA, CC, and THS, respectively) and on datasets from Monte Carlo simulations performed using the population PK model (n=50 x original group sizes) for 24-hour urine samples and for spot urine samples at 6 AM and 6 PM time points. Discrimination was conducted based on $CEMA_{creat}$ at Day 5.

Comparison	AUC_{ROC} (95% CI) 24h urine – Observed data	AUC_{ROC} (95% CI) 24h urine – Simulation	AUC_{ROC} (95% CI) Spot urine 6 AM – Simulation	AUC_{ROC} (95% CI) Spot urine 6 PM – Simulation
SA vs. CC	0.98 (0.96, 0.99)	0.93 (0.93, 0.94)	0.77 (0.76, 0.78)	0.97 (0.96, 0.97)
THS vs. CC	0.97 (0.96, 0.99)	0.94 (0.94, 0.95)	0.81 (0.81, 0.82)	0.97 (0.96, 0.97)

Table 4: ROC AUC estimates for observed and model simulated $CEMA_{creat}$ values in 24-hour and spot urine.

Discrimination Performance SA vs. CC – 24h Urine

Data	Accuracy %	Specificity (1-FPR) %	Sensitivity (TPR) %
Observed	93 (89, 95)	93 (89, 97)	92 (88, 96)
Simulated	87 (86, 87)	91 (90, 91)	83 (82, 84)

Table 5: Point and 95% interval estimates of discrimination performance for SA vs. CC using **cut-off=30 ng/mg creat**.

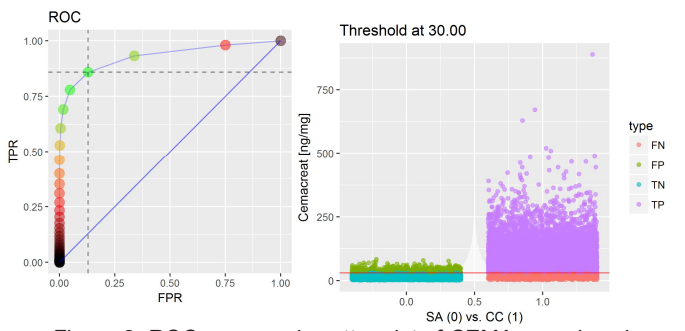
ROC
TPR
FPR
Threshold at 30.00
Cemacreat [ng/mg]
SA (0) vs. CC (1)
type
FN
FP
TN
TP

Figure 3: ROC curve and scatter plot of $CEMA_{creat}$ values in simulated 24-hour urine data.

Discrimination Performance THS vs. CC – 24h Urine

Data	Accuracy %	Specificity (1-FPR) %	Sensitivity (TPR) %
Observed	93 (91, 95)	94 (91, 96)	92 (88, 96)
Simulated	90 (90, 90)	94 (93, 94)	83 (82, 84)

Table 6: Point and 95% interval estimates of discrimination performance for THS vs. CC using **cut-off=30 ng/mg creat**.

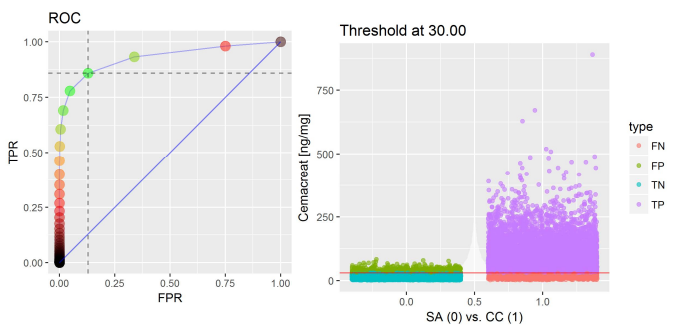
ROC
TPR
FPR
Threshold at 30.00
Cemacreat [ng/mg]
SA (0) vs. CC (1)
type
FN
FP
TN
TP

Figure 4: ROC curve and scatter plot of $CEMA_{creat}$ values in simulated 24-hour urine data.

Discrimination Performance THS vs. CC – 24h Urine

Data	Accuracy %	Specificity (1-FPR) %	Sensitivity (TPR) %	FPR %	FNR (1-TPR) %	DLR+	DLR-
Observed	93 (91, 95)	98 (96, 99)	84 (78, 89)	2 (1, 4)	16 (11, 22)	37.5 (20.1, 103.1)	0.2 (0.1, 0.2)
Simulated	90 (89, 90)	98 (98, 98)	74 (73, 75)	2 (2, 2)	26 (25, 27)	39.5 (35.3, 44.4)	0.27 (0.26, 0.28)

Table 7: Point and 95% interval estimates of detailed discrimination performance for THS vs. CC using **cut-off=40 ng/mg creat**.

Discrimination Performance SA vs CC – Spot Urine

Data	Accuracy %	Specificity (1-FPR) %	Sensitivity (TPR) %
Simulated @6AM	71 (70, 71)	83 (83, 84)	59 (58, 60)
Simulated @6PM	90 (90, 90)	99 (99, 100)	81 (80, 82)

Table 8: Point and 95% interval estimate of discrimination performance for SA vs. CC using **cut-off=6 ng/mg creat**.

Discrimination Performance THS vs CC – Spot Urine

Data	Accuracy %	Specificity (1-FPR) %	Sensitivity (TPR) %
Simulated @6AM	79 (79, 79)	90 (89, 90)	59 (58, 60)
Simulated @6PM	93 (93, 94)	99.54 (99.42, 99.64)	81 (80, 82)

Table 9: Point and 95% interval estimate of discrimination performance for THS vs. CC **cut-off=6 ng/mg creat**.

Acronyms: ROC=Receiver Operating Characteristic, FPR=False Positive Rate, TPR=True Positive Rate, DLR+/ DLR- positive/negative diagnostic likelihood ratio, TP=True Positive, FP= False Positive, TN=True Negative, FN=False Negative, Accuracy = (TP+TN)/(TP+TN+FP+FN)

Conclusions

The qualification of $CEMA_{creat}$ as a diagnostic biomarker provided an effective way to estimate users' integrated exposure to acrylonitrile and discriminate between smokers and subjects switching exclusively to THS or abstinence after five days, with no need of supportive information related to smoking behavior or number and type of products consumed. For spot urine assessments, highest discrimination performances of $CEMA_{creat}$ are observed in evening samples

Bibliography

1. Rodgman A, Perfetti TA. The chemical components of tobacco and tobacco smoke. 2nd ed: CRC Press, Taylor & Francis Inc (United States); 2013
2. Celerion. Unpublished on file data - Validation of a LC-MS/MS Method for the Determination of 3-HPMA, HBMA, and CEMA in Human Urine. 22 Oct 2015. Celerion Study ZZ36536-01. Lincoln, NE. 2015
3. ClinicalTrials.gov Identifiers: NCT-01959932, NCT-01970982, NCT-01970995, and NCT-01989156

SRNT 25th Annual Meeting, San Francisco, CA, USA, February 20–23, 2019

Competing Financial Interest – The research described in this poster was sponsored by Philip Morris International.