A meta-analysis of human organotypic respiratory cultures exposed to whole cigarette smoke

P. Leroy, C. Mathis, M. Cabanski, A. Iskandar, R. Kostadinova, S. Frentzel, D. Kuehn, S. Majeed, C. Merg, A. Elamin, E. Guedj, R. Dulize, Y. Xiang, F. Martin, W. Schlage, M.C. Peitsch, J. Hoeng

Philip Morris International Research and Development, Philip Morris Product SA, Quai Jeanrenaud 5, CH-2000 Neuchâtel, Switzerland

INTRODUCTION

The development and the use of human three-dimensional *in vitro* models that closely mimic *in vivo* biology are of great interest to the scientific community to overcome some of the limitation of species translatability and further support the reduction, refinement, and replacement framework of animal use in laboratory. Human organotypic epithelial tissue cultures (e.g., bronchial, nasal or buccal), grown at the air-liquid interface, are particularly attractive to assess the impact of airborne toxicants exposure, including pollutants and cigarette smoke (CS).

As organotypic tissue models are currently still expensive and not always accessible for small laboratories, having a well-defined experimental design is essential for scientifically sound outcomes. We investigated the 'reproducibility' of different molecular endpoints collected after CS exposure of human organotypic bronchial and nasal epithelial cultures obtained from different production batches as well as from different donors.

The following endpoints were assessed after different post-exposure time points of 28 min exposure to air (Sham control) or to different dilutions of mainstream whole smoke (3R4F – Health Canada regimen): cytotoxicity (Adenyl Kinase assay), cytochrome P450 (CYP1A1/1B1, CYP3A4) activity, pro-inflammatory markers secretion (MAP).The statistical analysis shows the impact and the contribution of the use of various donors as well as multiple exposure smoke experiments.

MATERIALS & METHODS

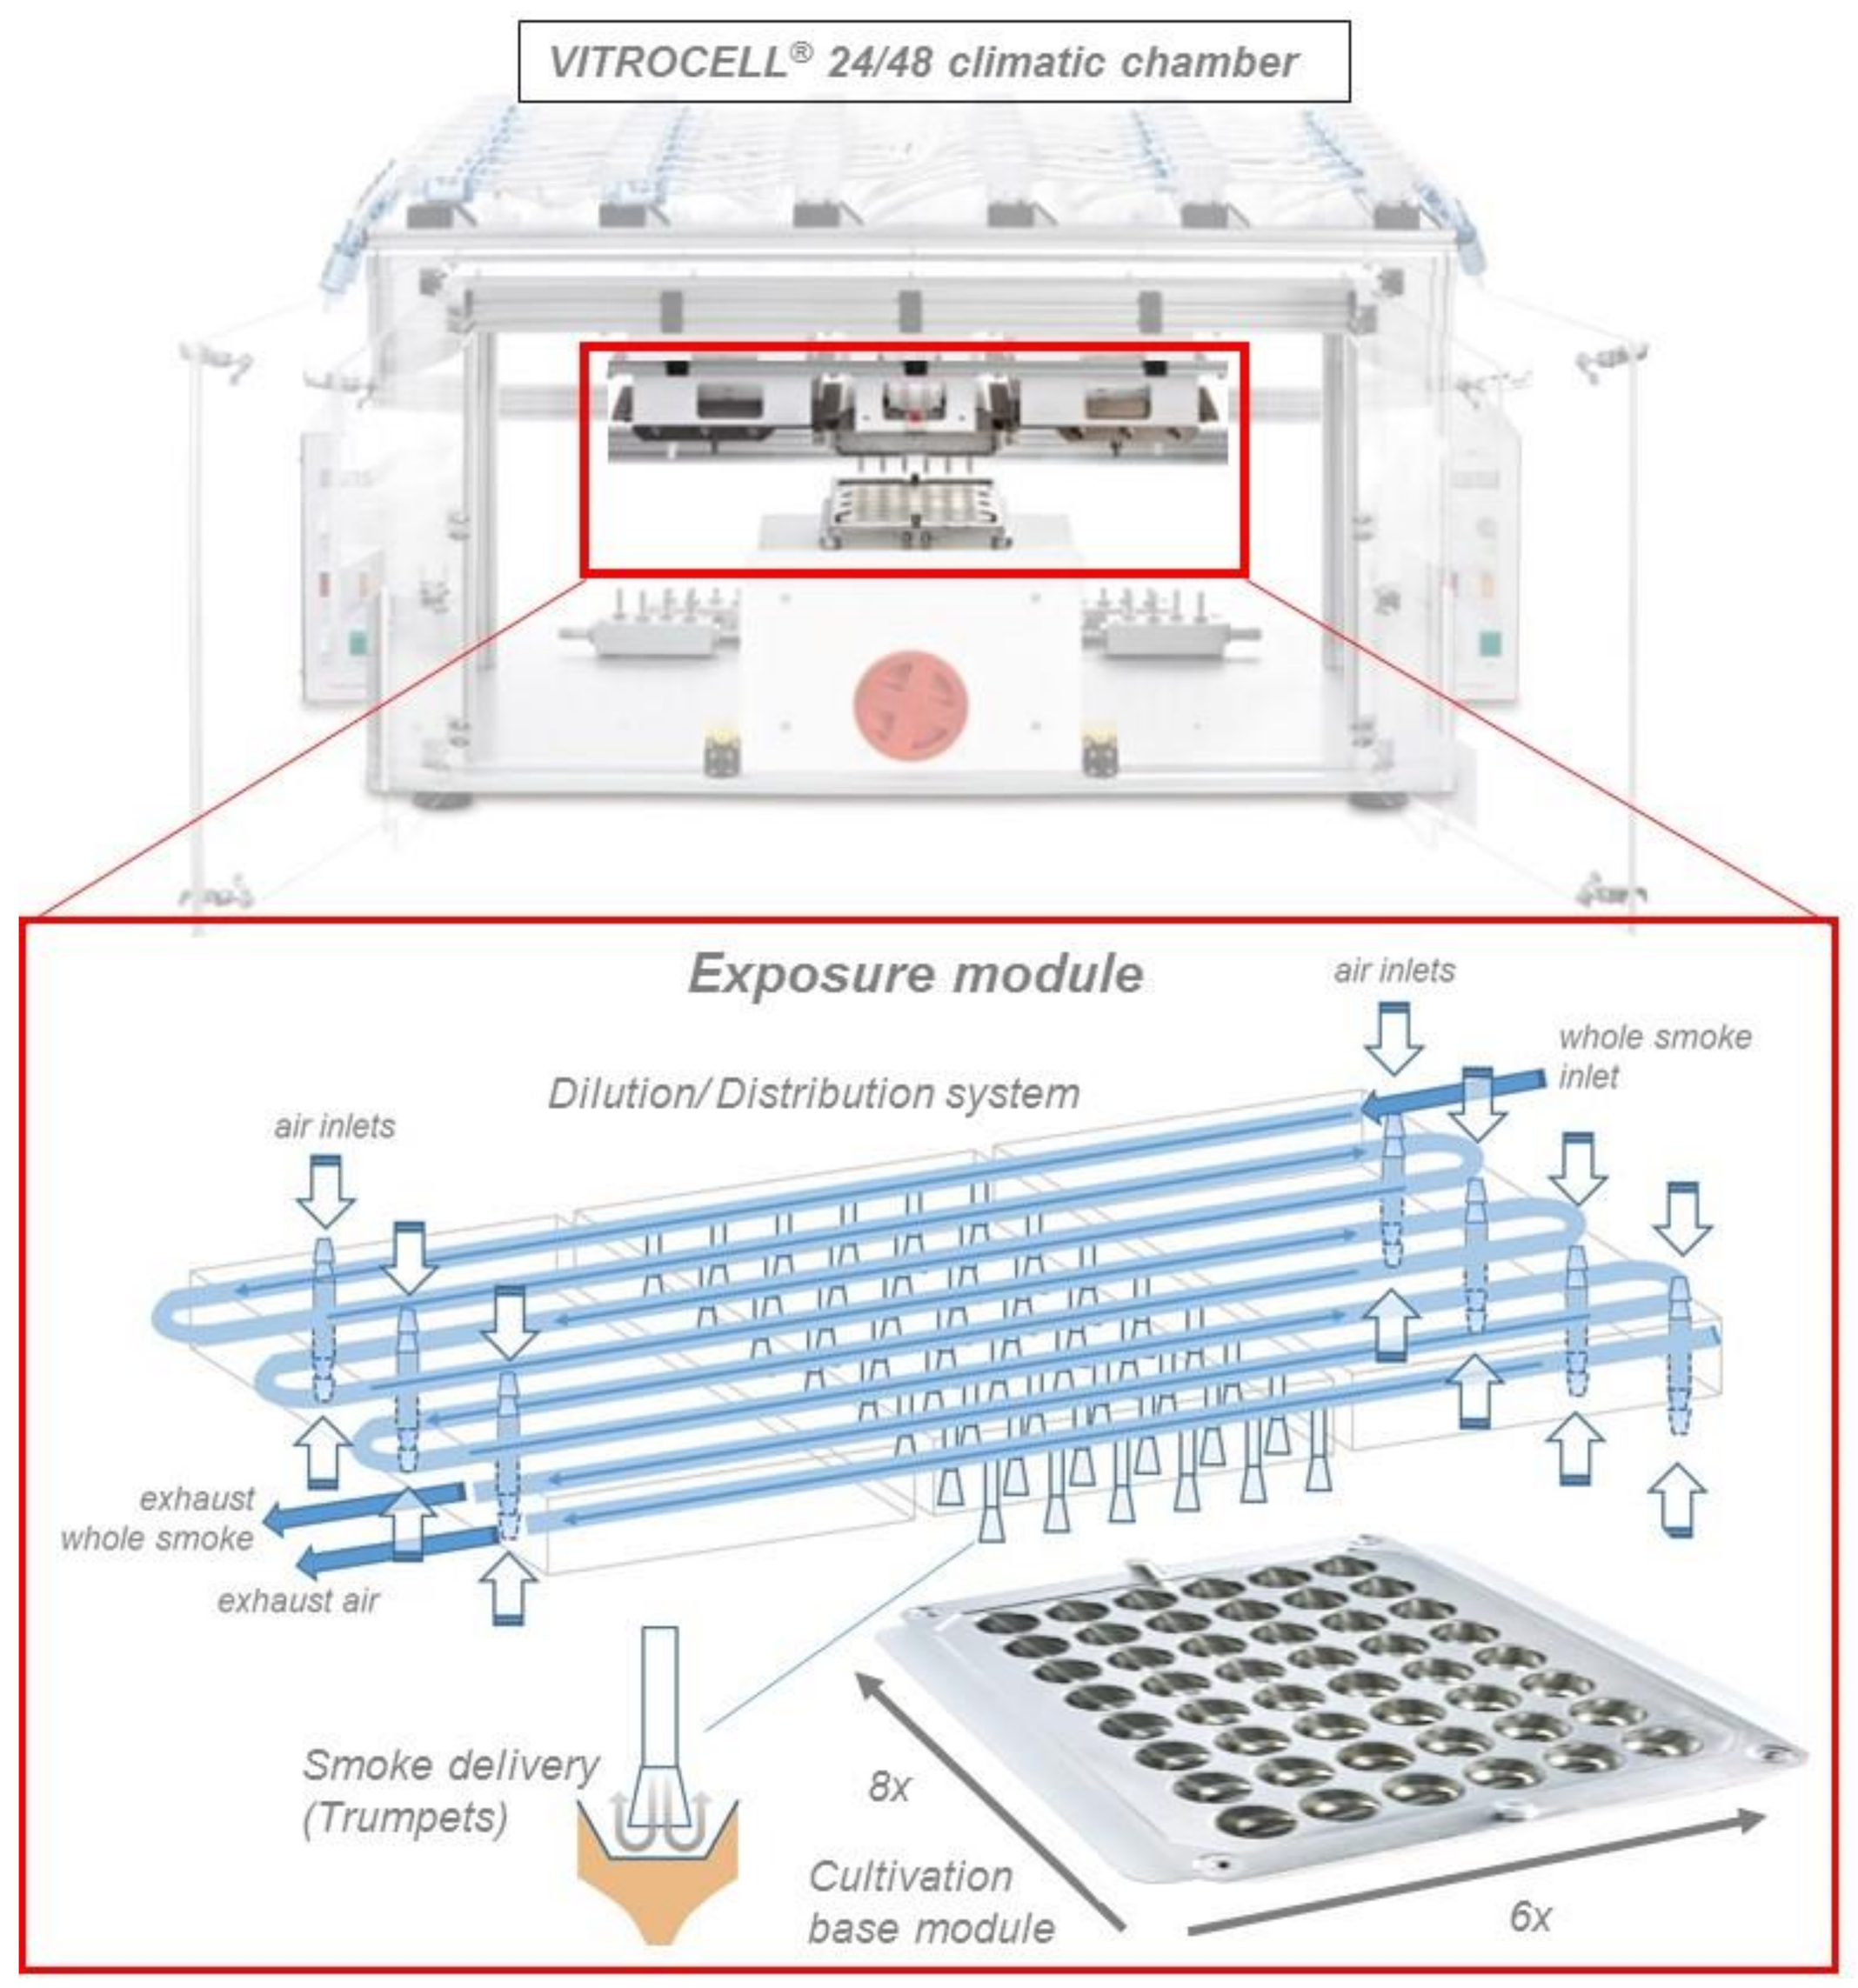
VITROCELL® 24/48 climatic chamber
Exposure module
air inlets
whole smoke inlet
Dilution/Distribution system
air inlets
exhaust whole smoke
exhaust air
Smoke delivery (Trumpets)
8x
Cultivation base module
6x

Figure 1: Schematic representation of the VITROCELL® 48 exposure system.

Human organotypic epithelial tissue cultures (purchased from Epithelix SARL or MatTek®) are exposed in parallel to humidified air (control) or to diluted mainstream CS (reference cigarette 3R4F, University of Kentucky) with Health Canada smoking regimen adapted to Vitrocell® exposure system.

A climatic chamber houses an exposure module, which consists of a Dilution/ Distribution system, on top of a Cultivation base module. In the base module, up to 48 wells can be exposed simultaneously. The base module has a format of 8 rows x 6 columns.

The delivery of whole smoke is achieved by individual trumpets, delivering the smoke from the Dilution/ Distribution system to the wells of the Cultivation base module.

Additional QCM (not shown) can be connected to each row of the Dilution/ Distribution system, to monitor particle deposition.

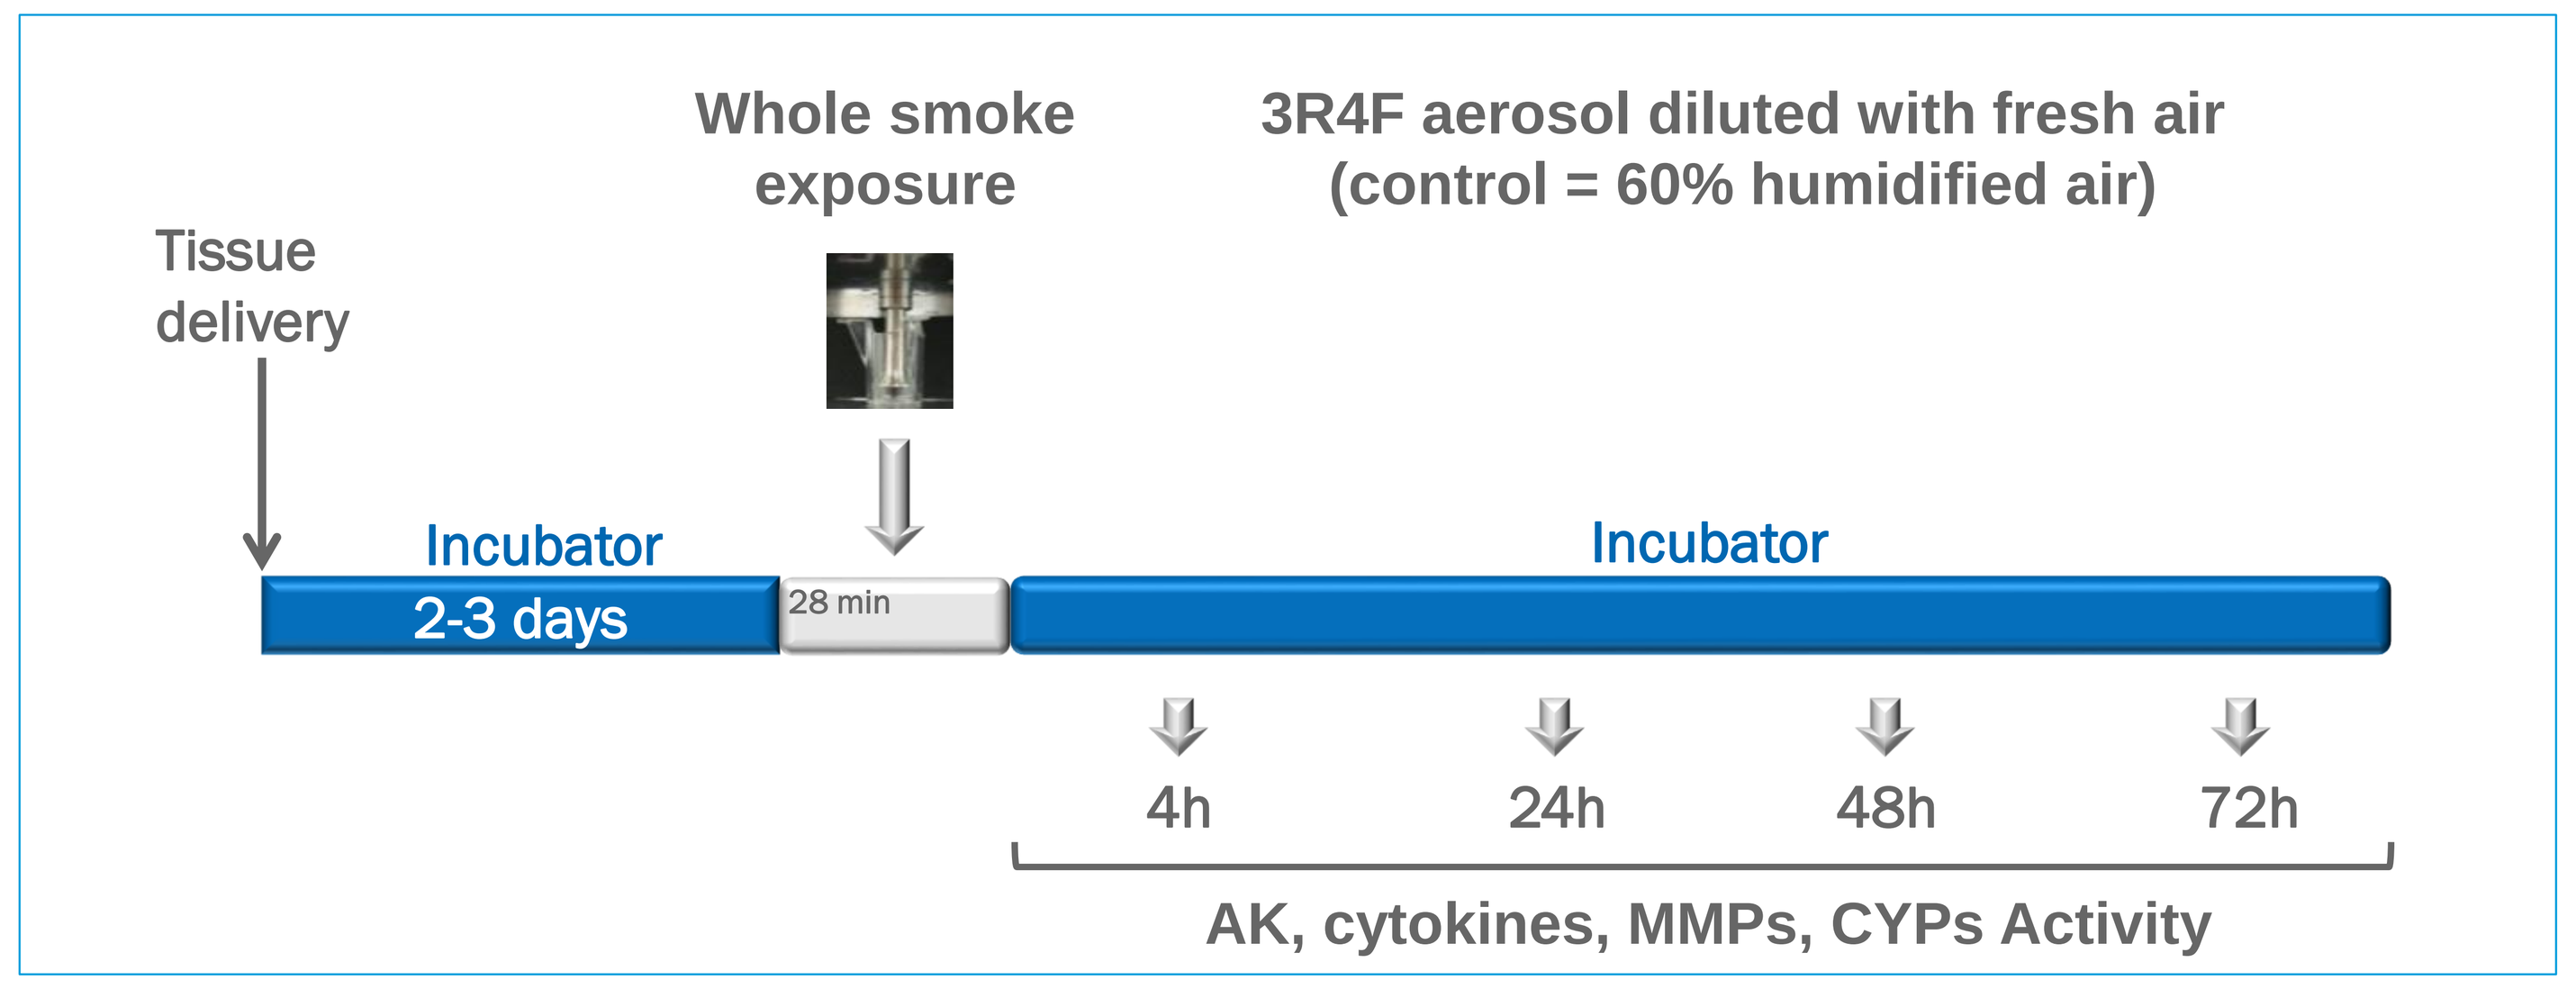
Whole smoke exposure
3R4F aerosol diluted with fresh air (control = 60% humidified air)
Tissue delivery
Incubator
2-3 days
28 min
Incubator
4h
24h
48h
72h
AK, cytokines, MMPs, CYPs Activity

Figure 2: *Before and after the exposure, the tissues are kept at 37℃ in the incubator. Then measurement of different endpoints at various post-exposure time points were performed:*

- ***AK** using the* Promega's *ADP-Glo™ Kinase* Assay *and* ***CYP1A1/1B1*** *and* ***CYP3A4***, *using the Promega's P450-Glo™ Luminescent Cytochrome P450 Assay, on a BMG LABTECH OPTIMA FLUOstar® microplate reader,*
- ***Cytokines** and **chemokines** analysis on a Luminex® 200™ with various kits : HMMP2MAG-55K (MMP-1 and MMP-9), HCYTOMAG-60K (CCL11, CSF3, CSF2, IL1A, IL1B, IL6, CXCL8, CXCL10, CCL2, CCL5, VEGFA), HTMP2MAG-54K (EGF, CXCL1P, IL10, IL13, TIMP1, TNFA), HCYT_SEPSIS (CCL20, TSLP, sICAM1)*

DESIGN OF STUDIES

Tissue	Year	Calendar Week	Study ID	Smoke Run	Donor ID														Dose [%]									Post Exposure [h]			
				N	A	B	C	D	E	F	G	H	I	J	K	L	M	N	0	8	10	13	15	20	30	40	50	4	24	48	72
Bronchial without Fibroblast	2013	39	1	1	.	.	.	.	.	.	.	X	.	.	.	.	.	.	X	X	.	X	.	.	.	.	.	X	X	X	.
		40	2	1	.	.	.	.	.	.	.	X	.	.	.	.	.	.	X	X	.	.	X	.	.	.	.	X	X	X	X
		45	4	1	.	.	.	.	.	.	.	X	.	.	.	.	.	.	X	X	.	.	X	.	.	.	.	X	X	X	X
		49	5	2	.	.	.	.	.	.	.	X	.	.	.	.	.	.	X	X	.	.	X	.	.	.	.	X	X	X	X
	2014	8	6	3	.	.	.	.	.	.	.	X	.	.	.	.	.	.	X	X	.	.	X	.	.	.	.	X	X	X	X
		10	7	3	X	.	.	X	X	.	.	.	.	.	.	.	X	.	X	.	X	.	.	.	.	.	.	.	.	X	X
		17	8	6	.	.	.	X	.	.	.	.	.	.	.	.	.	.	X	X	.	.	X	.	.	.	.	X	.	X	.
Nasal without Fibroblast	2013	37	1	1	.	.	X	.	.	.	.	.	.	.	X	.	.	.	X	.	X	.	X	.	.	.	.	X	X	X	.
		47	2	1	.	.	.	.	.	.	.	.	.	X	.	.	.	.	X	X	.	.	X	.	.	.	.	.	X	X	.
		48	3	2	.	.	.	.	.	.	.	.	.	.	.	.	.	X	X	X	.	.	X	.	.	.	.	X	X	X	X
	2014	10	4	3	.	X	.	.	.	.	.	.	X	X	.	.	.	.	X	X	.	.	.	.	.	.	.	.	.	X	.
		16	5	3	.	.	.	.	.	.	.	.	X	.	.	.	.	.	X	X	.	.	X	.	.	.	.	X	X	X	X
		26	6	3	.	.	.	.	.	.	.	.	X	.	.	.	.	.	X	X	.	X	X	.	.	.	.	X	X	X	X
Buccal without Fibroblast	2014	19	1	3	.	.	.	.	.	X	X	.	.	.	.	.	.	.	X	.	X	.	.	X	X	X	X	.	X	X	.
		25	2	3	.	.	.	.	.	X	.	.	.	.	.	.	.	.	X	.	.	.	.	X	X	.	.	X	X	.	X
		27	3	3	.	.	.	.	.	X	.	.	.	.	.	.	.	.	X	.	.	.	.	X	X	.	.	X	X	.	X

Table 1. *Representation of studies with specific designs to address a variety of scientific questions such as:*

- *What is the maximal dose of cigarette smoke that can be delivered per tissue without inducing cytotoxicity?*
- *What is the optimal post-exposure time for the biological endpoints sample?*
- *What is the biological response of the respiratory cultures to whole cigarette smoke?*

The generate results from those of 16 studies has lead to a rich data compendium suitable for meta-analysis.

DONOR & SMOKE RUN EFFECT

Illustration of smoke run effect and donor effect on one representative cytokine CCL11.

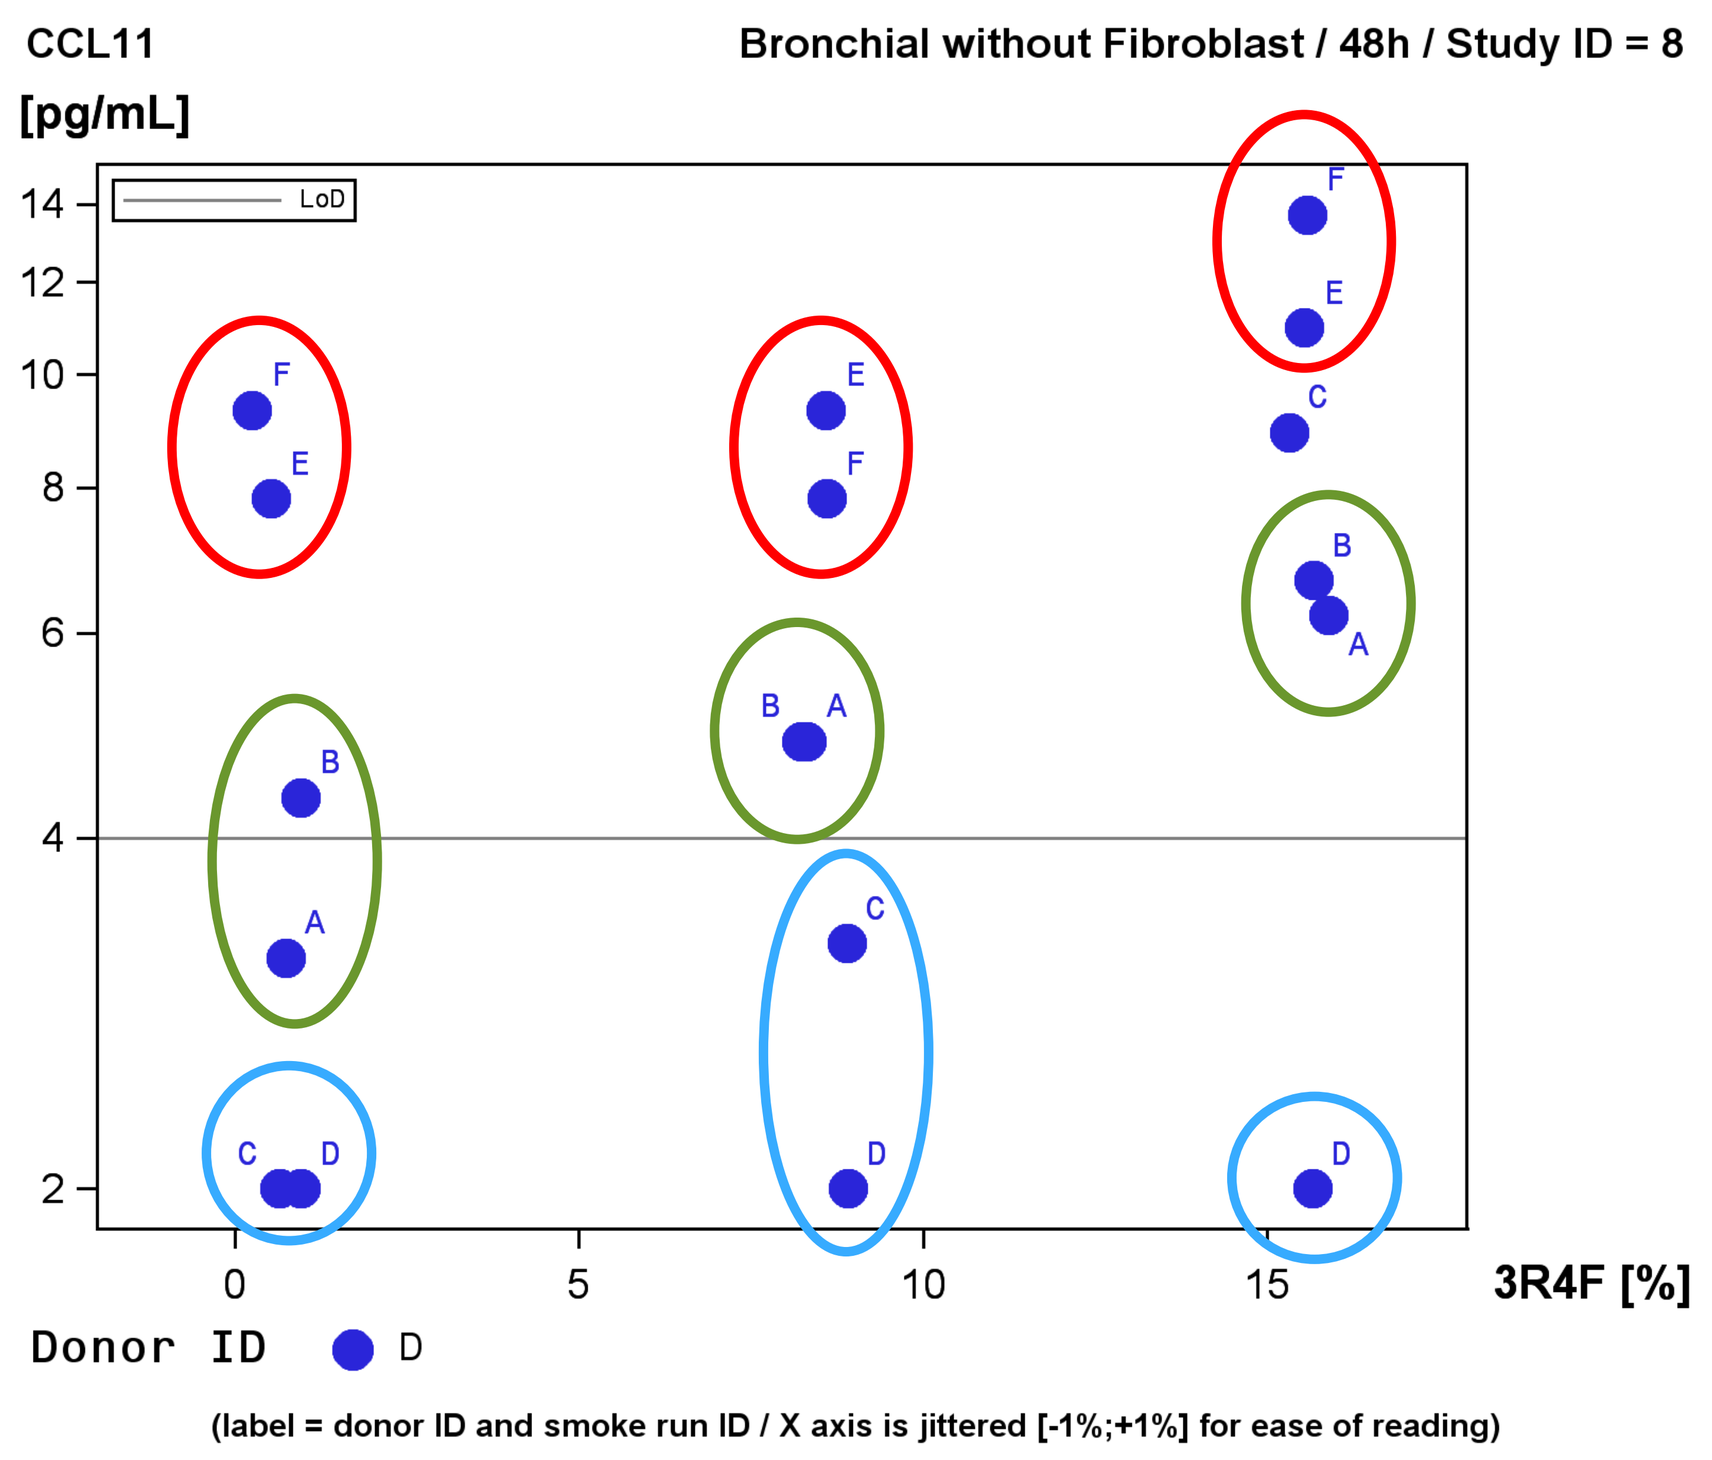
CCL11
[pg/mL]
Bronchial without Fibroblast / 48h / Study ID = 8
LoD
3R4F [%]
Donor ID D
(label = donor ID and smoke run ID / X axis is jittered [-1%;+1%] for ease of reading)

Figure 3: CCL11 abundance is higher in the experiments with *smoke runs* *E and F compared to smoke runs A and B or C and D. These effects are consistent across the 3R4F dilution range.*

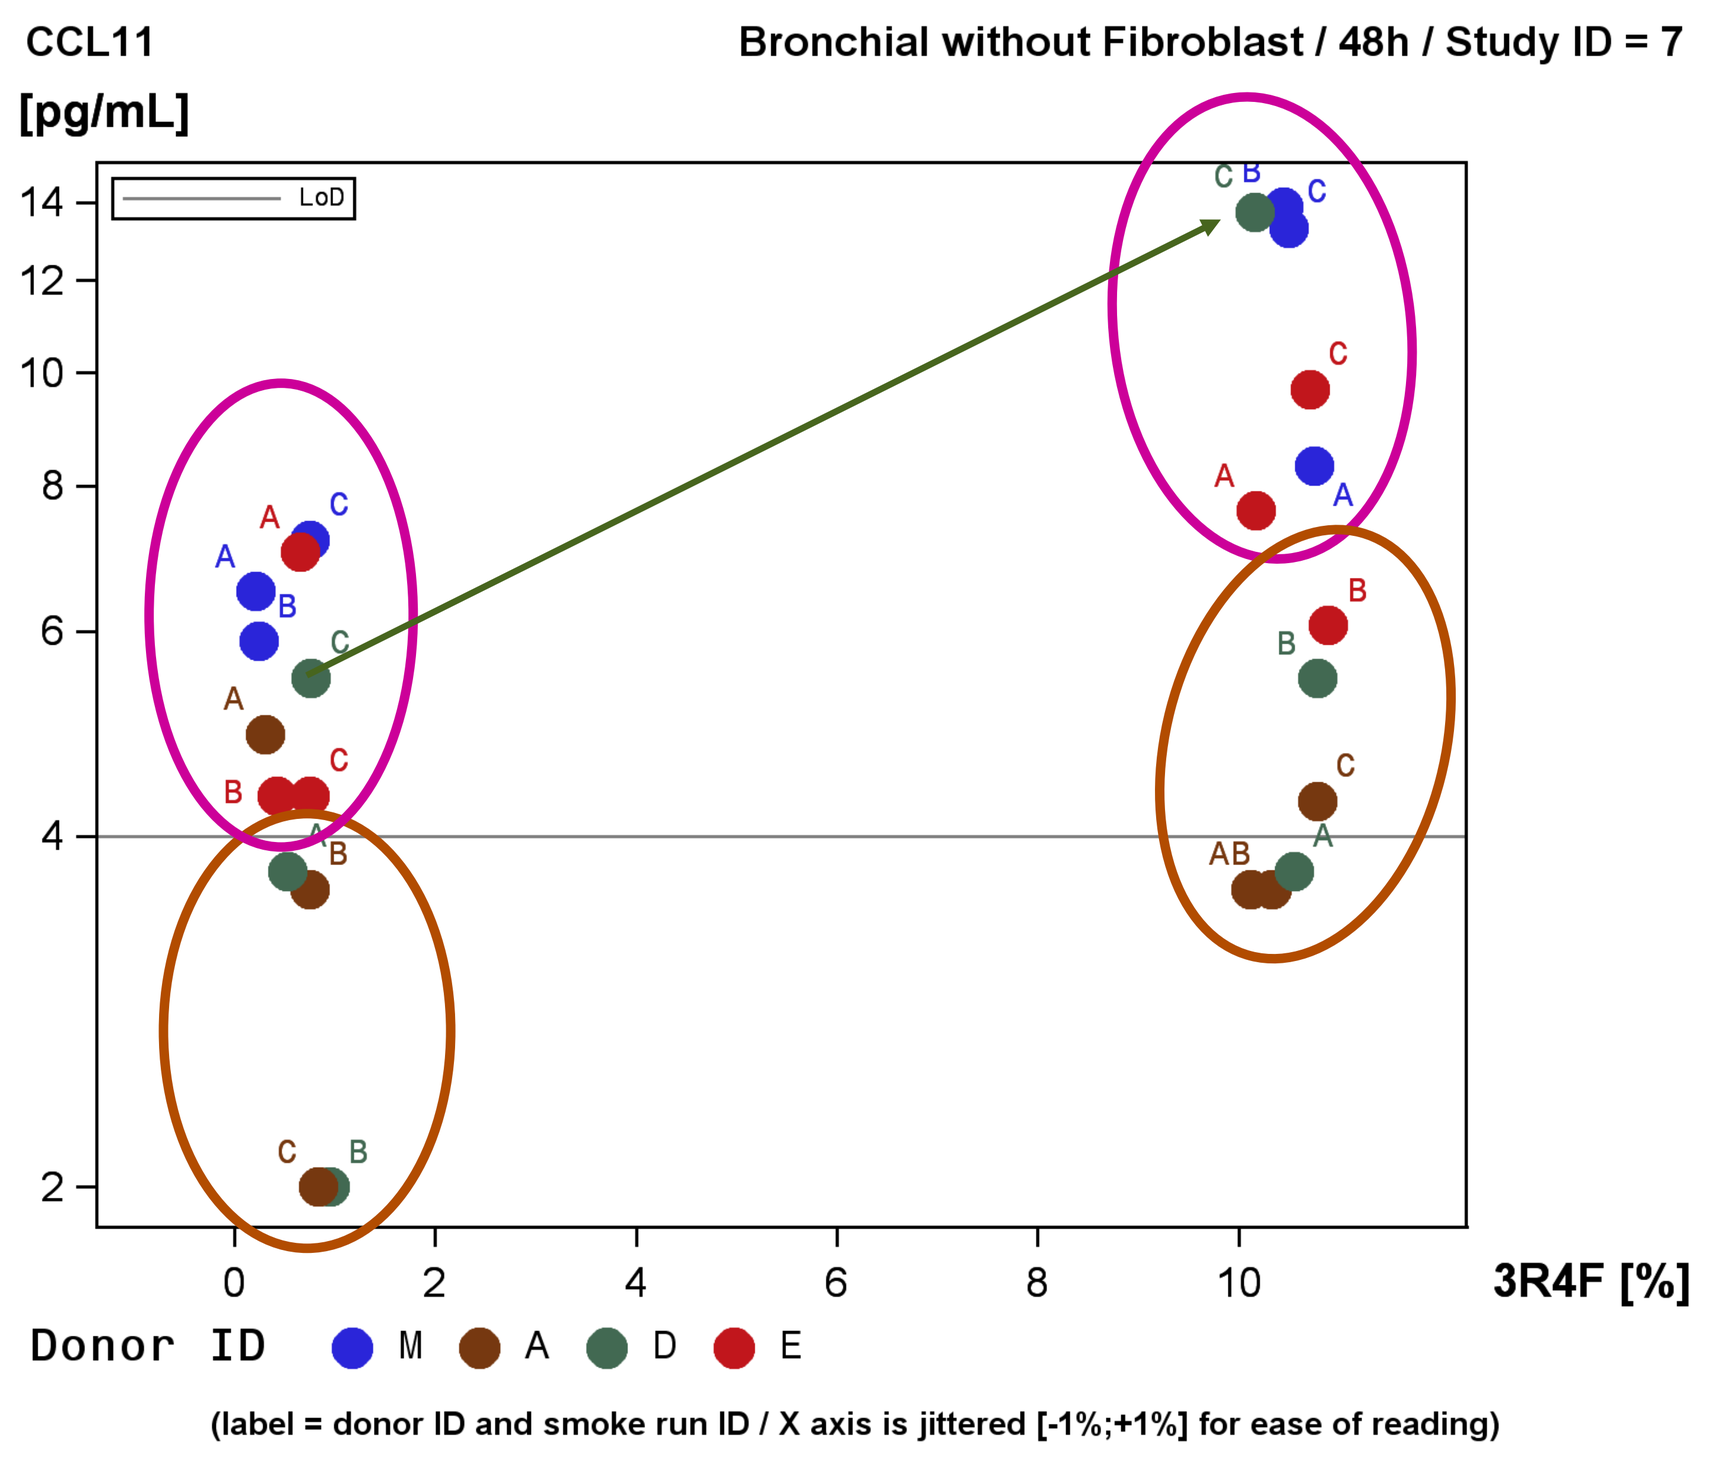
CCL11
[pg/mL]
Bronchial without Fibroblast / 48h / Study ID = 7
LoD
3R4F [%]
Donor ID M A D E
(label = donor ID and smoke run ID / X axis is jittered [-1%;+1%] for ease of reading)

Figure 4: CCL11 abundance is higher for donors M and E compared to donor A and D, thus the statistical model selected included the donor effect.

STATISTICAL MODEL

$$\log_{10}\Big(\underbrace{Y}_{\text{measured marker}}\Big) = \overbrace{\underbrace{X}_{\text{known fixed effect values}} * \underbrace{\beta}_{\text{unknown fixed effect parameters}}}^{\text{ANCOVA on smoke concentration}} + \overbrace{\underbrace{Z}_{\text{known design matrix}} * \underbrace{\gamma}_{\text{unknown random effect parameters}}}^{\text{Donor+Smoke Run correction}} + \underbrace{\varepsilon}_{\text{Gaussian random errors}}$$

Figure 5: Overview of the statistical model. A MIXED model has been fitted on the log-transformed endpoint values. The model is based on an ANCOVA (a blend of ANOVA and regression over the different smoke concentration), and takes into account a correction for the different donors and smoke runs.

RESULTS OF THE STATISTICAL ANALYSIS

Figure 6

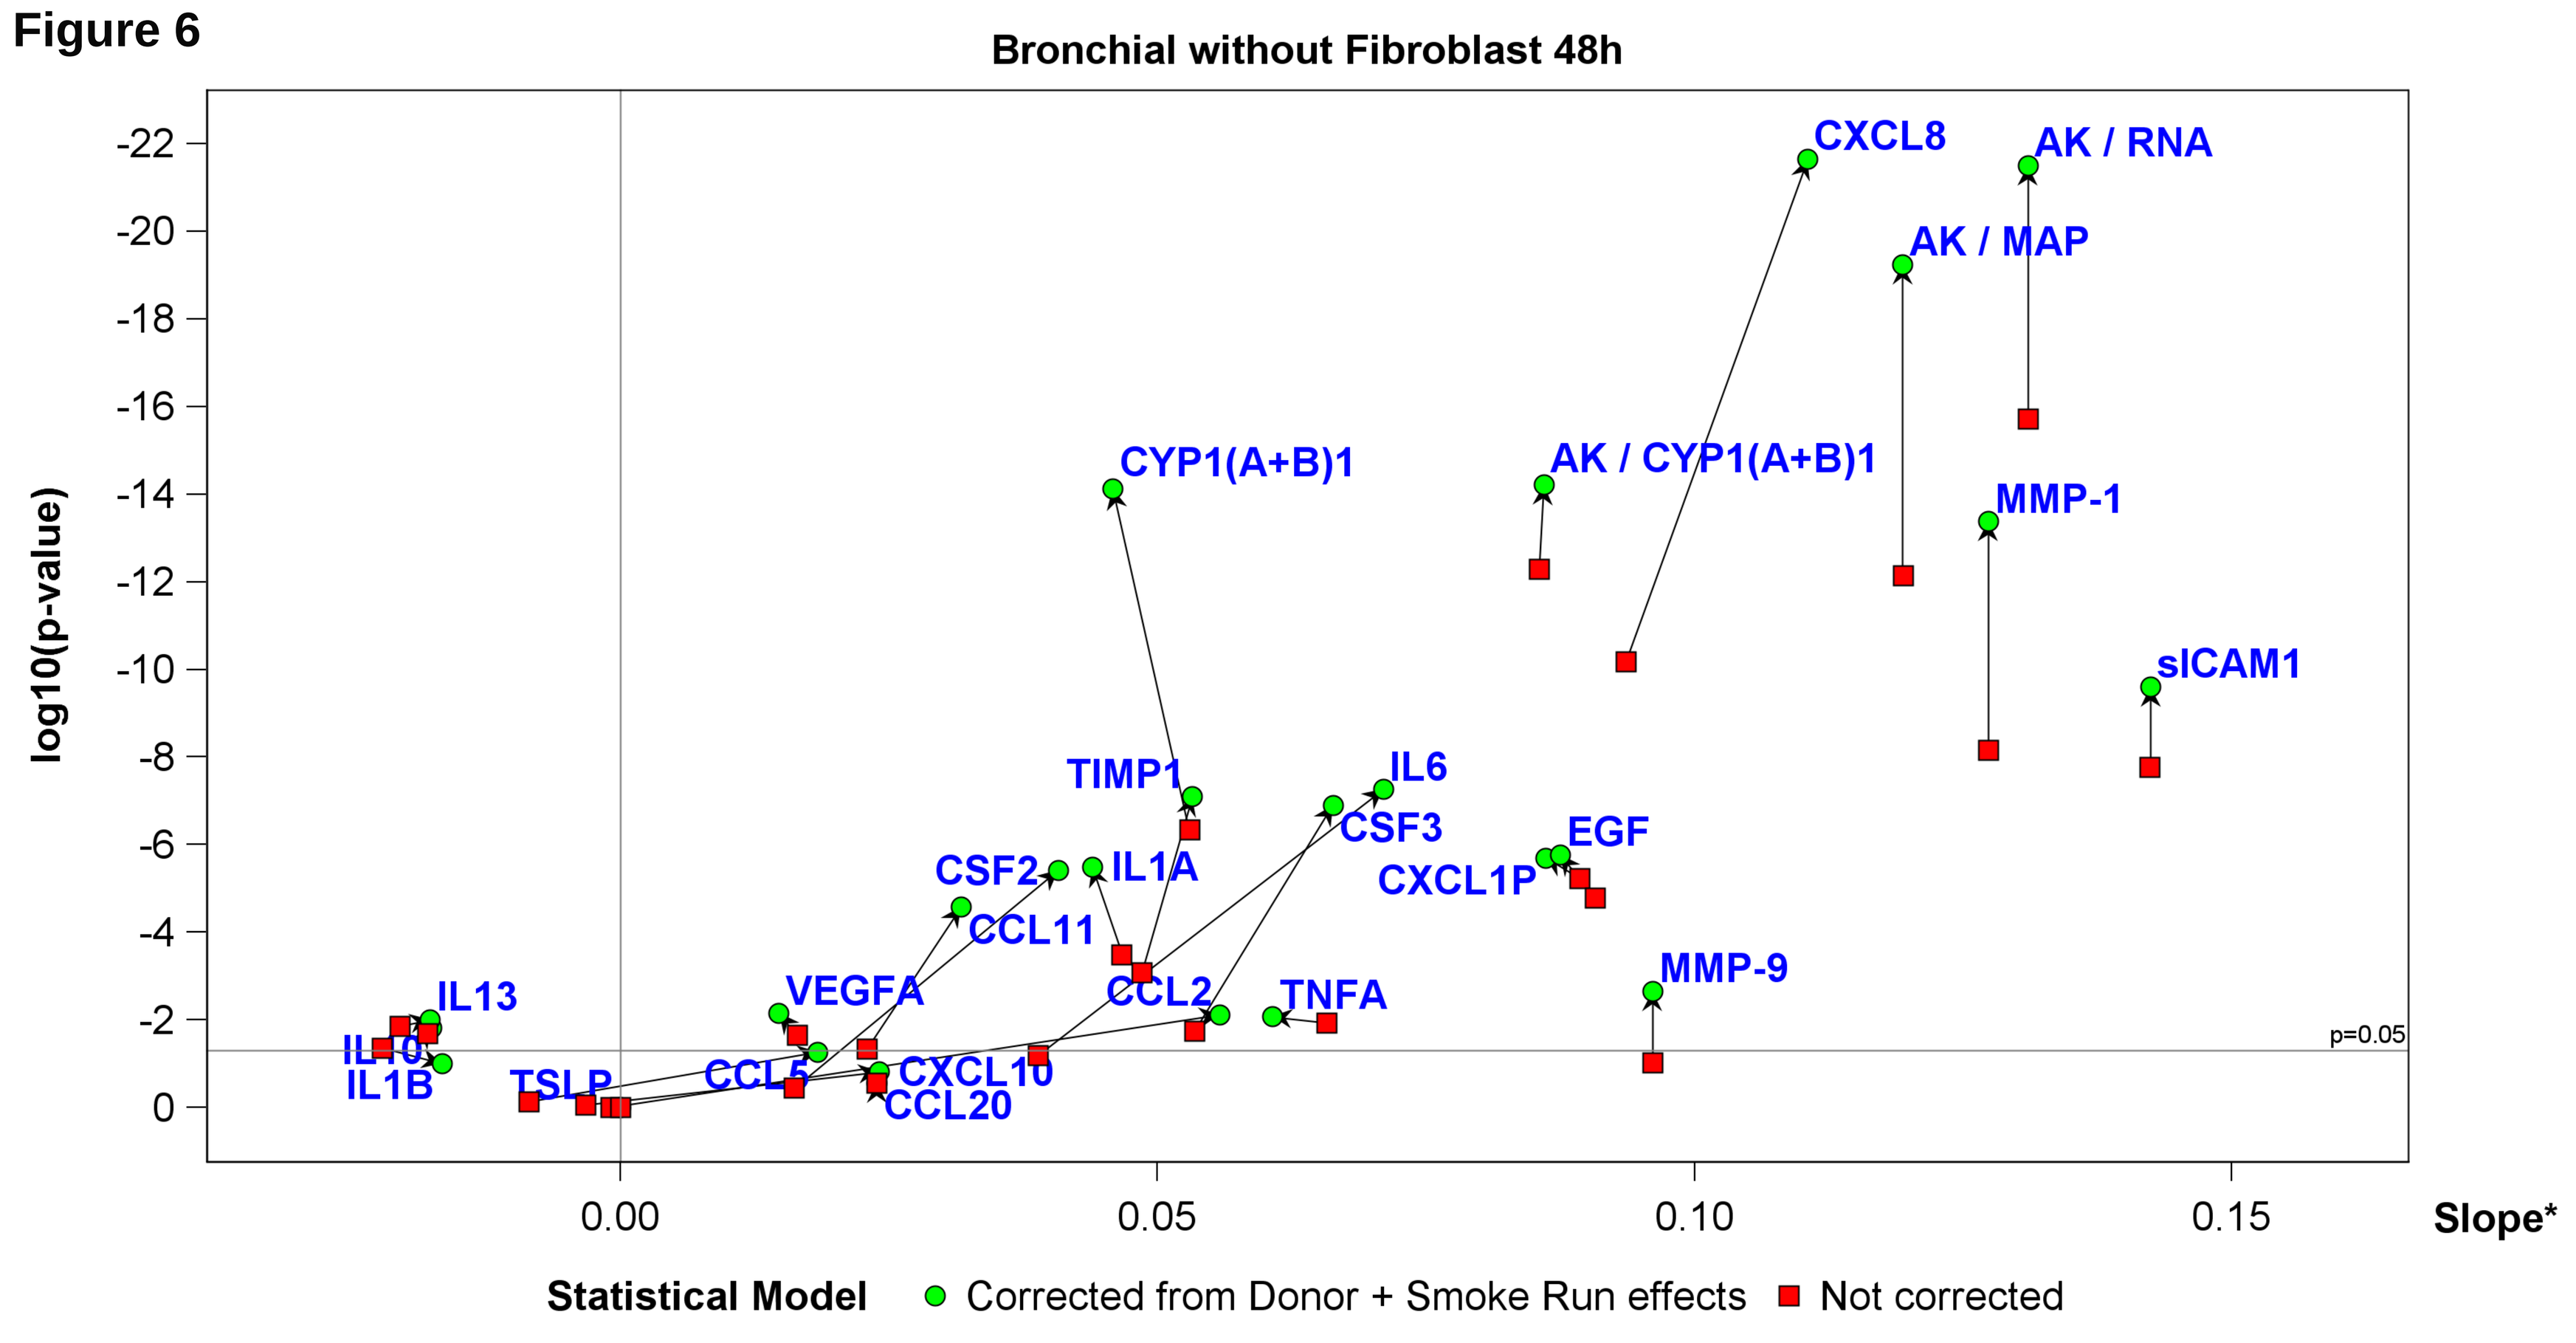
Bronchial without Fibroblast 48h
log10(p-value)
CXCL8
AK / RNA
AK / MAP
CYP1(A+B)1
AK / CYP1(A+B)1
MMP-1
sICAM1
TIMP1
IL6
CSF3
EGF
CXCL1P
CSF2
IL1A
CCL11
MMP-9
IL13
VEGFA
CCL2
TNFA
IL10
IL1B
TSLP
CCL5
CXCL10
CCL20
p=0.05
Slope*
Statistical Model
Corrected from Donor + Smoke Run effects
Not corrected

Figure 7

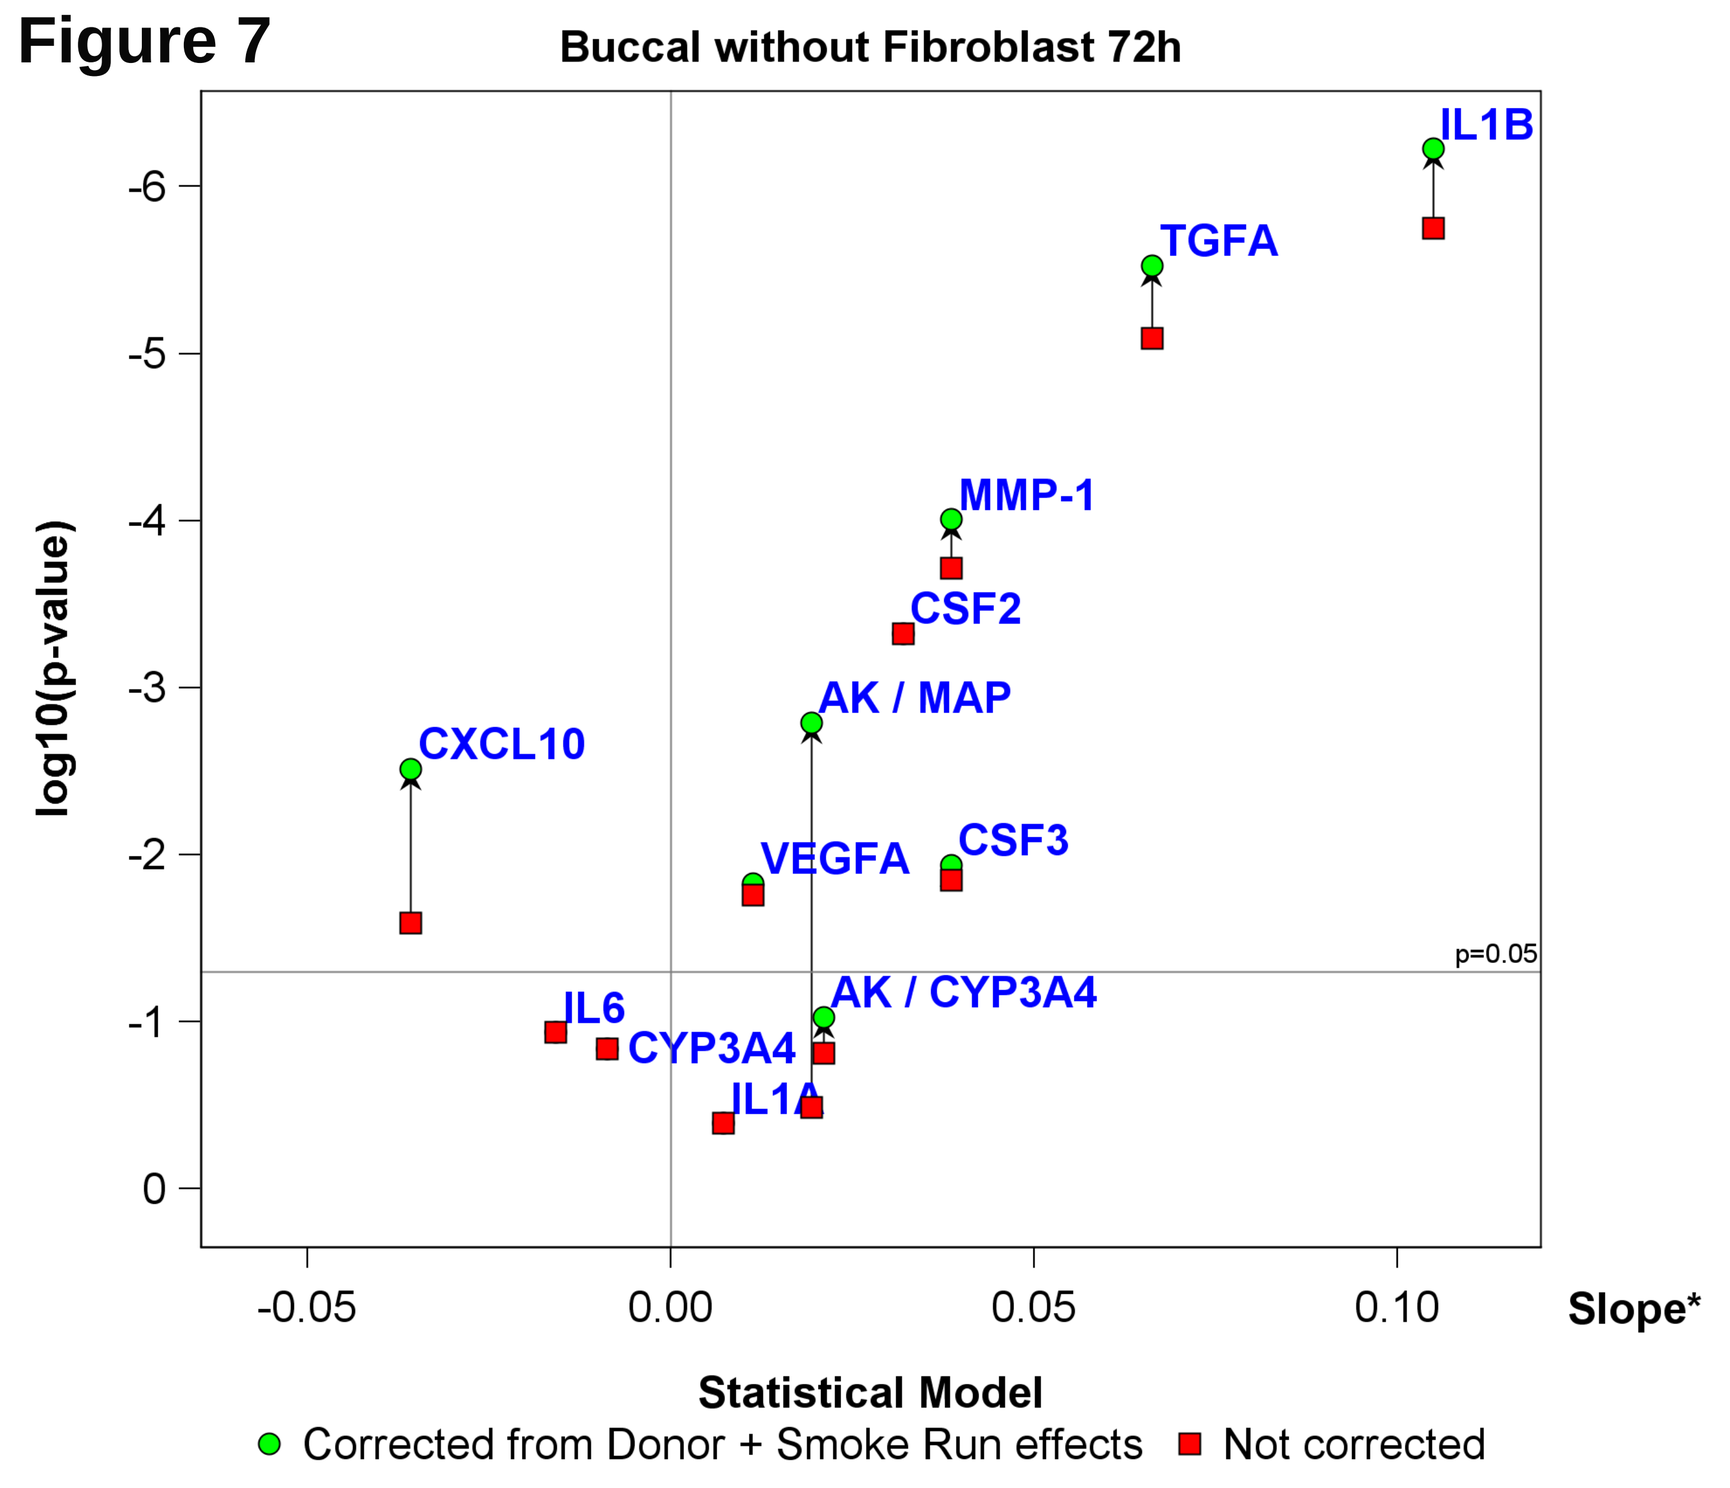
Buccal without Fibroblast 72h
log10(p-value)
IL1B
TGFA
MMP-1
CSF2
AK / MAP
CXCL10
VEGFA
CSF3
IL6
CYP3A4
AK / CYP3A4
IL1A
p=0.05
Slope*
Statistical Model
Corrected from Donor + Smoke Run effects
Not corrected

Figure 8

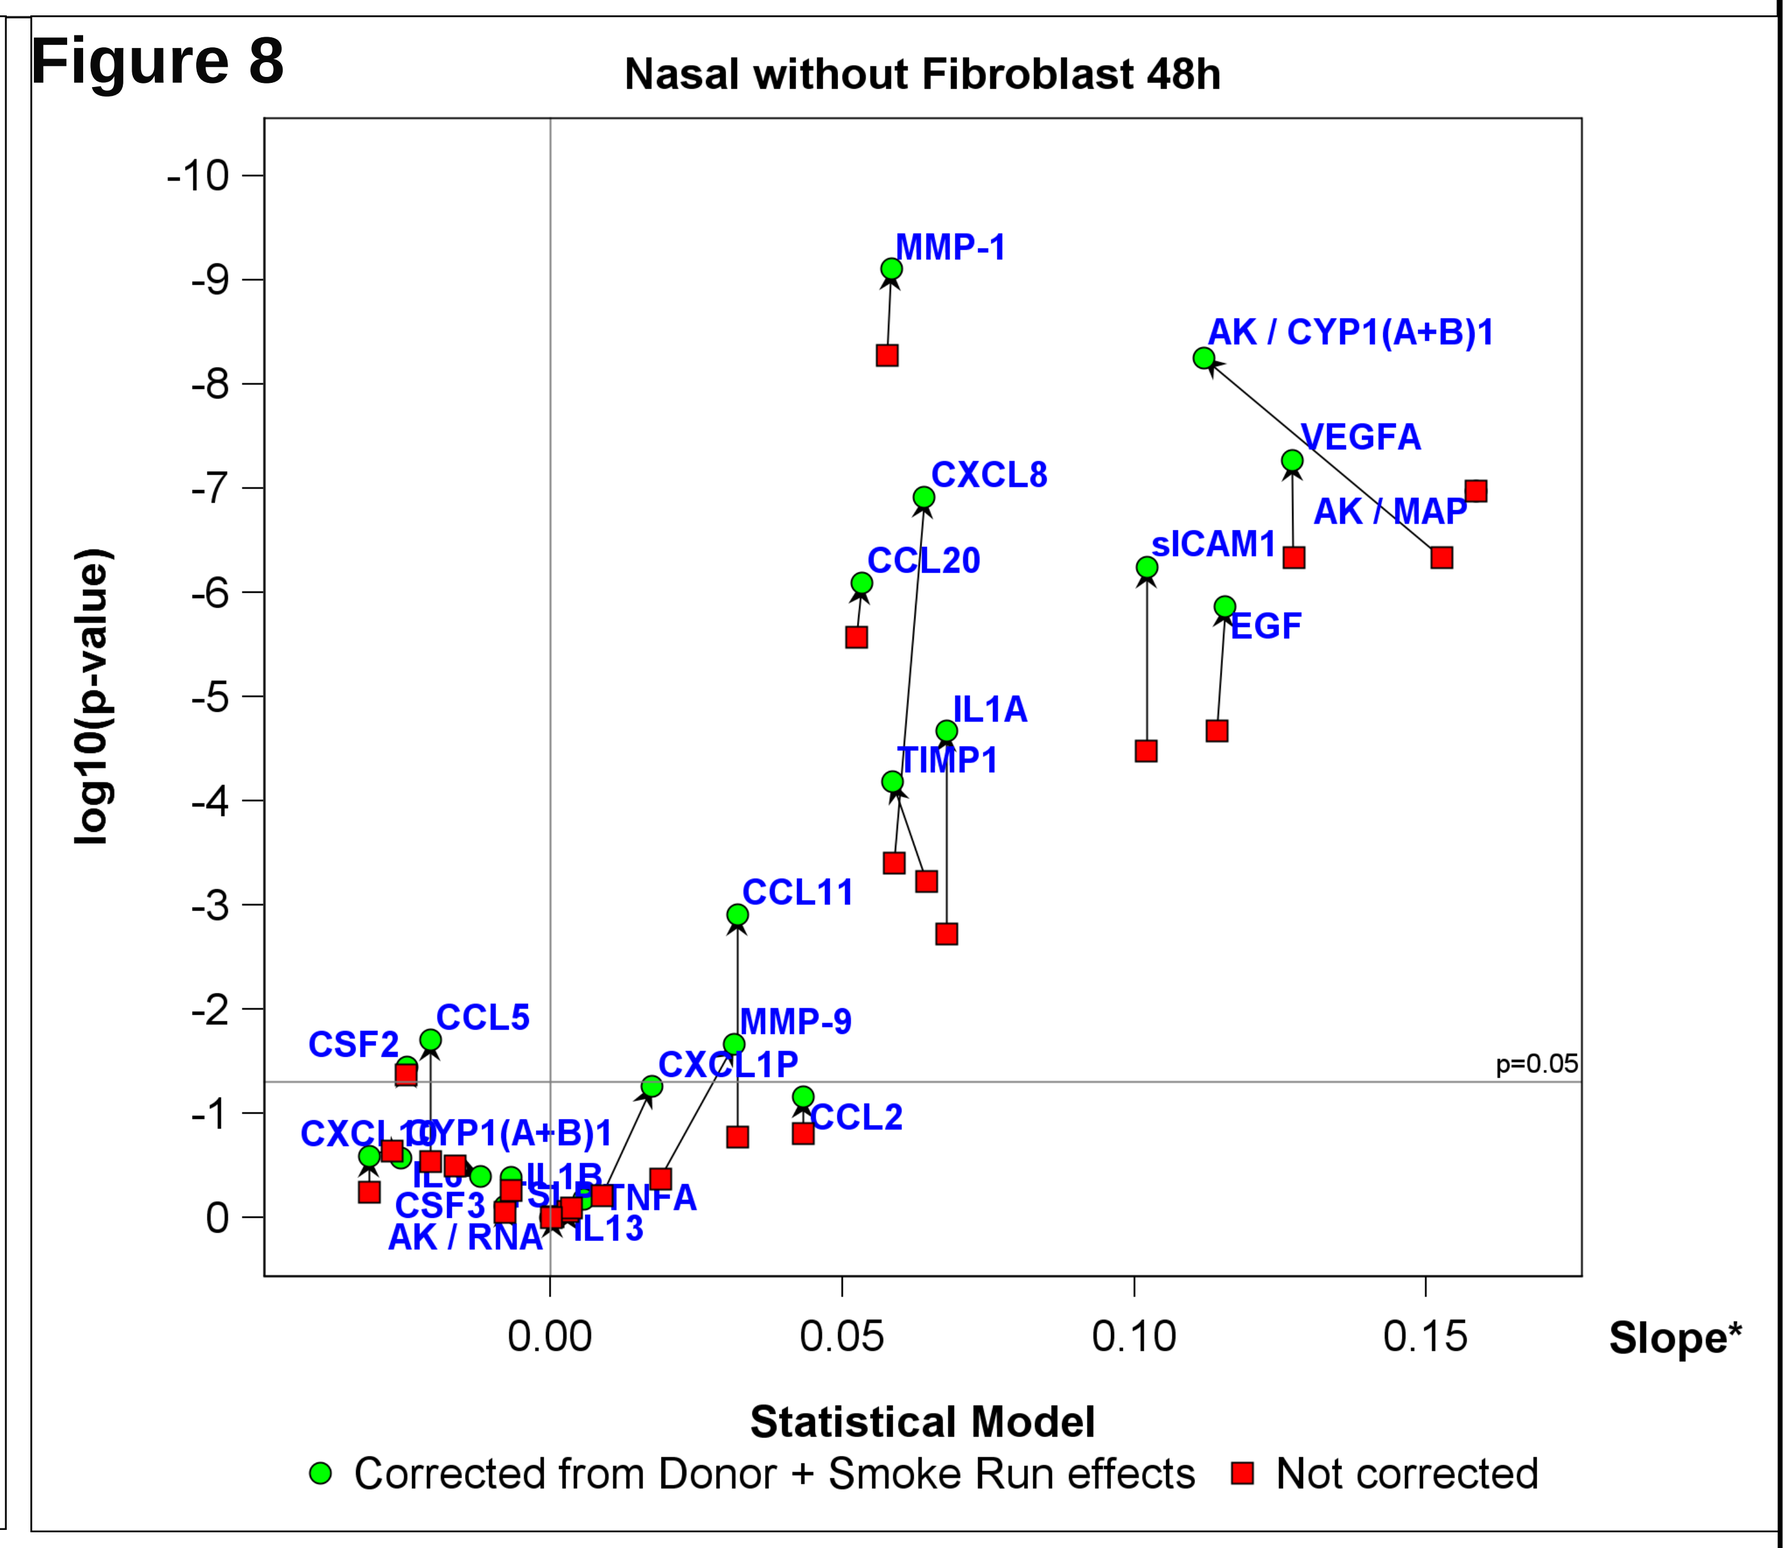
Nasal without Fibroblast 48h
log10(p-value)
MMP-1
AK / CYP1(A+B)1
VEGFA
CXCL8
AK / MAP
CCL20
sICAM1
EGF
IL1A
TIMP1
CCL11
CSF2
CCL5
MMP-9
CXCL1P
CCL2
TNFA
IL13
p=0.05
Slope*
Statistical Model
Corrected from Donor + Smoke Run effects
Not corrected

Figure 6,7, and 8: Overview of the difference between a model corrected for donor and smoke run effects and non-corrected, for various endpoints across the three organotypic culture models

- Upward arrows represent that the corrected model better fits the data resulting in smaller p-values
- Vertical arrows indicate that effect size stays consistent between corrected and non-corrected model.

(* Slope of 0.05, 0.10, 0.15 translate respectively to +15%, +25%, +41% increase by % of smoke concentration.)

DISCUSSION / CONCLUSIONS

- While usually the biological replicate is considered as the experimental unit, in this meta-analysis the smoke run was treated as experimental unit in the statistical model. When multiple tissue inserts were utilized within one smoke run for endpoint determination, then the median of the endpoint values was included in the statistical model thereby ensuring that:
 - All smoke runs have the same weight in the statistical model avoiding overrepresentation and consequently bias.
 - P-values are not artificially lowered by increasing the number of biological replicates.
 - Results are more likely to be robust and reproducible when repeated
- From the biological perspective a dose response can be observed for the majority of pro-inflammatory in all three organotypic tissue models when exposed to whole cigarette smoke; similarly CYP activities and AK values respond in a dose dependent manner .
- The statistical model developed for the meta-analysis provides enhanced correction with increasing number of smoke runs and donors for the endpoints of interest.

REFERENCES

1. **Human bronchial epithelial cells exposed in vitro to cigarette smoke at the air-liquid interface resemble bronchial epithelium from human smokers.** *C. Mathis, et al*. Am J Physiol Lung Cell Mol Physiol. 2013 Apr 1;304(7):L489-503.

PMI RESEARCH & DEVELOPMENT

2014 ACT Annual Meeting – November 9-12, 2014 – Orlando

Philip Morris International Research & Development,
Quai Jeanrenaud 5, 2000 Neuchâtel, Switzerland
T: +41 58 242 21 13, **F**: +41 58 242 28 11, **W**: www.pmi.com