Systems toxicology analysis of cardiovascular and respiratory endpoints from ApoE$^{-/-}$ mice showed similar effects when switching to a candidate Modified Risk Tobacco Product, THS2.2, or ceasing smoking

Phillips B.[1], Guedj E.[2], Elamin A.[2], Boue S.[2], Vuillaume G.[2], Martin F.[2], Leroy P.[2], Sierro N.[2], Hayes A.W.[3], Veljkovic E.[1], Ivanov N.V.[2], Peitsch M.C.[2] and Hoeng J.[2]

[1]Philip Morris International Research Laboratories, 50 Science Park Road, 117406 Singapore (Part of Philip Morris International group of companies) [2]Philip Morris International R&D, Philip Morris Products S.A., Quai Jeanrenaud 5, 2000 Neuchâtel, Switzerland (Part of Philip Morris International group of companies) [3]Spherix Consulting, Rockville, MD, USA and Scientific Advisory Board PMI R&D

Background

Smoking causes many serious diseases including cardiovascular disease (CVD) and chronic obstructive pulmonary disease (COPD). Suitable animal models play an important role in understanding of smoke-induced pathogenesis. This study examined the development of hallmarks of both COPD and CVD in ApoE$^{-/-}$ mice exposed to either cigarette smoke (CS) or to an aerosol from a candidate modified risk tobacco product (MRTP), the tobacco heating system (THS2.2) over an 8-month period. In addition to chronic exposure regimes, a comparison of exposure cessation or switching to THS2.2 after 2 months of exposure to CS was performed using a battery of assays (physiological, morphological and molecular). Biological interpretation of various endpoints enables to understand the biological effects of cessation/switching as compared to continuous smoking.

Study design and endpoints

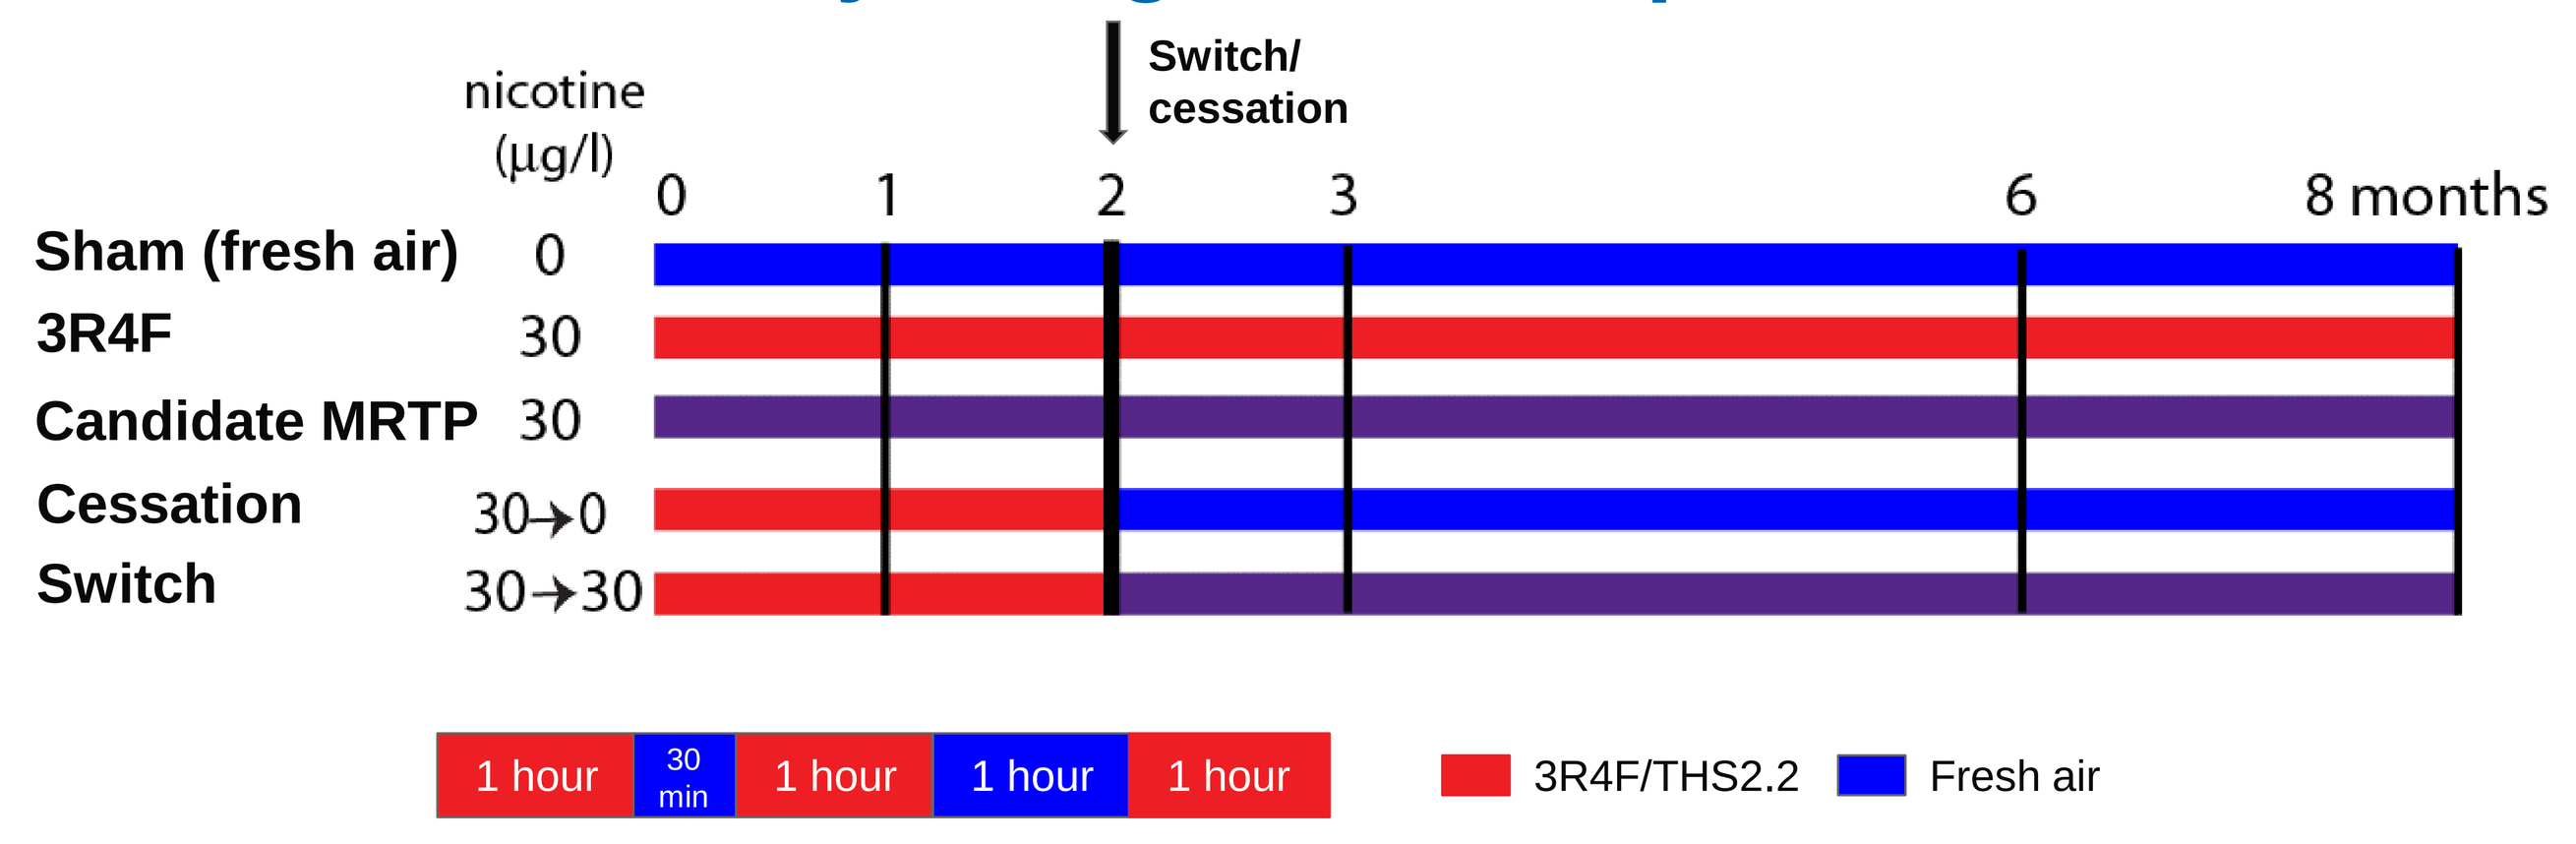
nicotine (µg/l)
Switch/ cessation
0 1 2 3 6 8 months
Sham (fresh air) 0
3R4F 30
Candidate MRTP 30
Cessation 30→0
Switch 30→30
1 hour
30 min
1 hour
1 hour
1 hour
3R4F/THS2.2
Fresh air

Female ApoE$^{-/-}$ mice were exposed to 3R4F (600 mg/m^3 TPM), THS2.2 (matched to the nicotine in 3R4F – 30 µg/l) or filtered air for 3 hours per day, 5 days per week, for up to 8 months. After 2 months of exposure to 3R4F, switching and cessation groups were exposed to aerosol from THS2.2 or filtered air, respectively. Animals were observed on a daily basis, body weight progression was monitored twice per week, exposure parameters (carboxyhemoglobin (COHb) in blood and nicotine metabolites in urine) were measured 3 times during the study. Dissections were performed after 1, 2, 3, 6, and 8 months of exposure. At each time point animals were allocated for the following endpoints: bronchoalveolar lavage fluid (BALF), identification of infiltrated inflammatory cells in lungs and multi-analyte (cytokines/chemokines) profiling; histopathological evaluation and morphometry of lungs; lung function; plaque surface determination and an extensive molecular high throughput analysis (transcriptomics, proteomics and lipidomics).

Exposure biomarkers

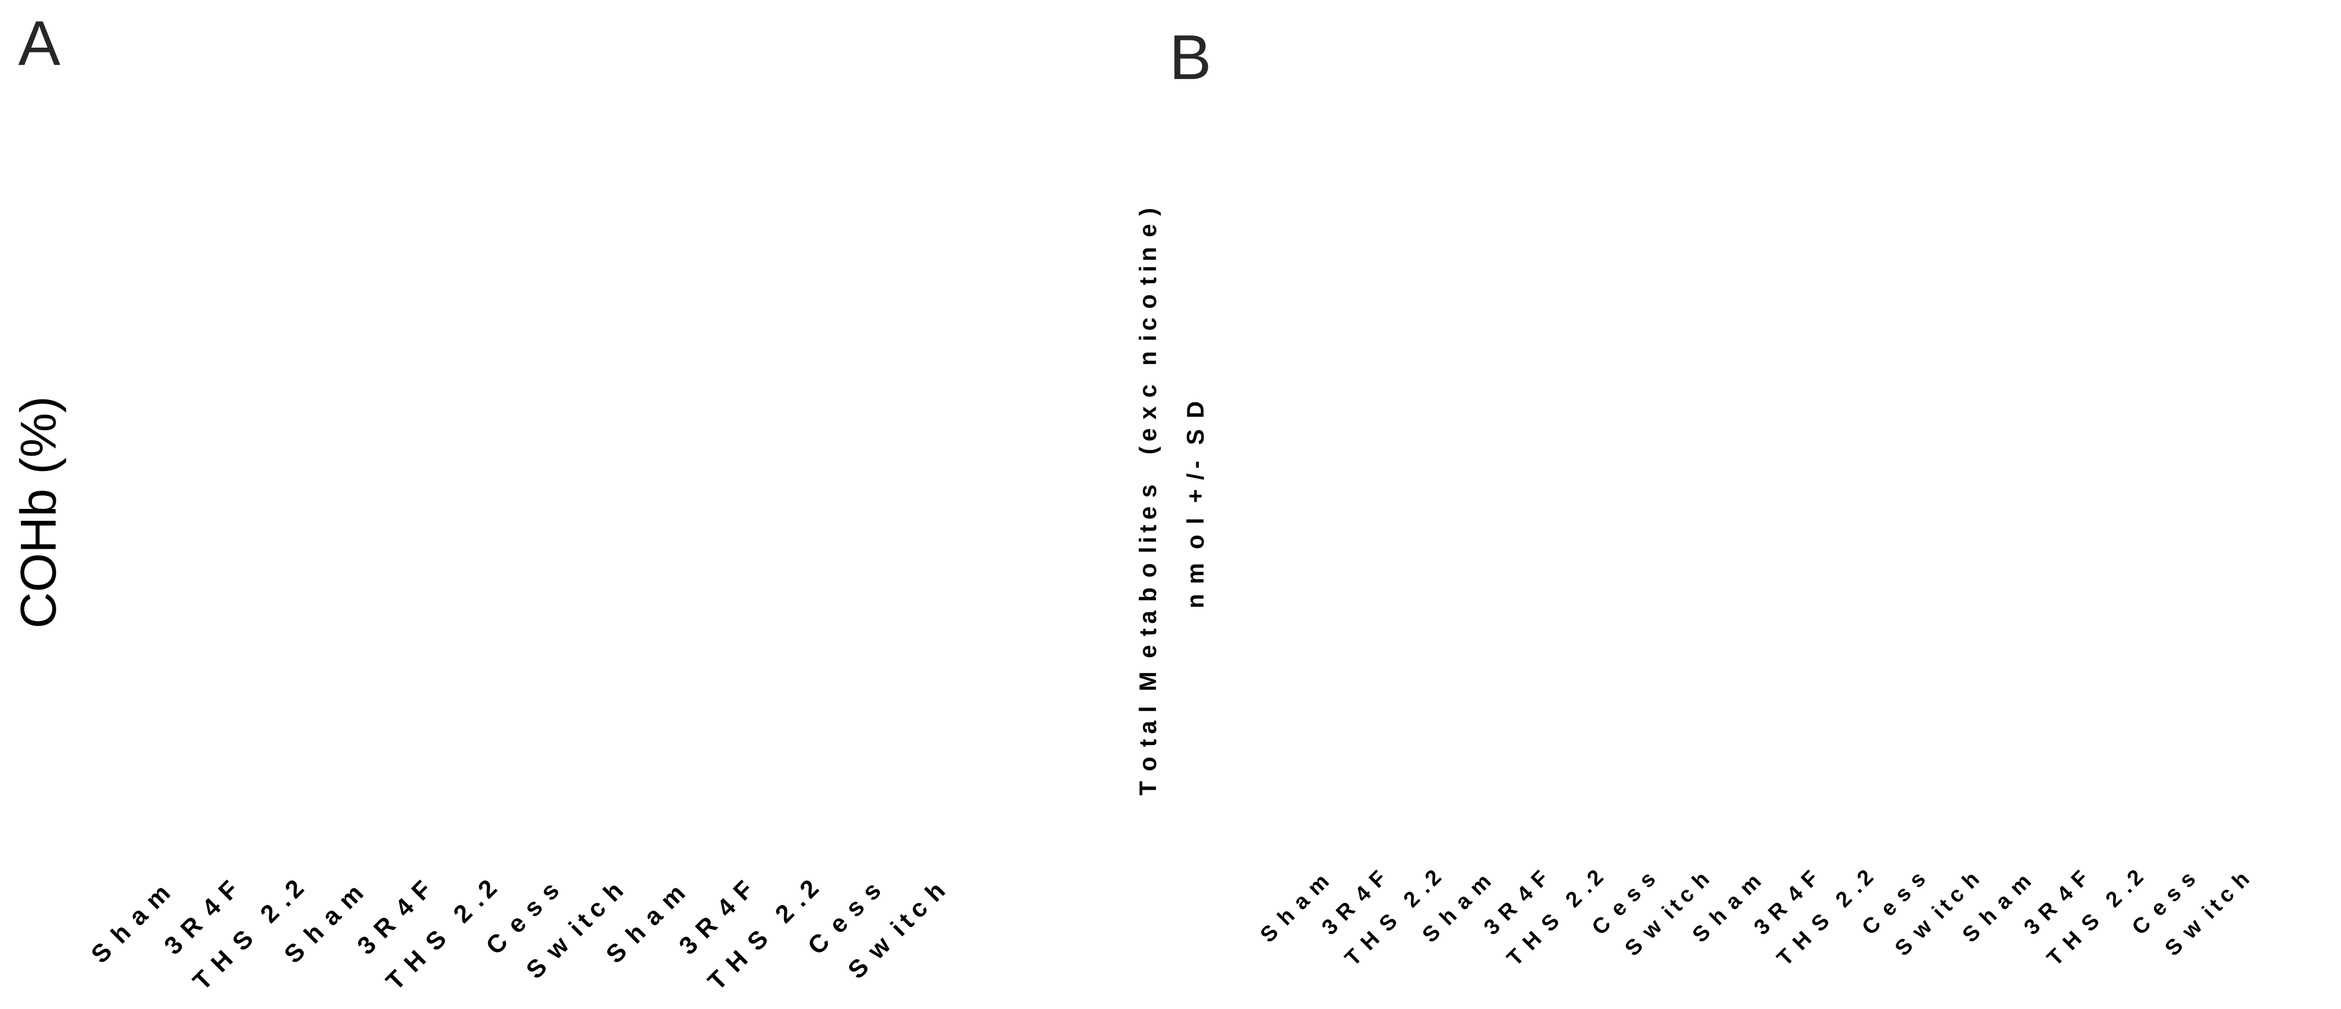
A
B
COHb (%)
Total Metabolites (exc nicotine) nmol +/- SD

After switching to the THS2.2 aerosol or filtered air, levels of COHb in blood (A) of exposed animals were substantially lower compared to those found in samples of mice continuously exposed to 3R4F. Measured concentration of nicotine metabolites in urine (B) (3'-hydroxycotinine, norcotinine, cotinine, nicotine-N'-oxide and nornicotine) confirmed that mice received equivalent nicotine concentrations in the different groups exposed to 3R4F or THS2.2.

Lung inflammation

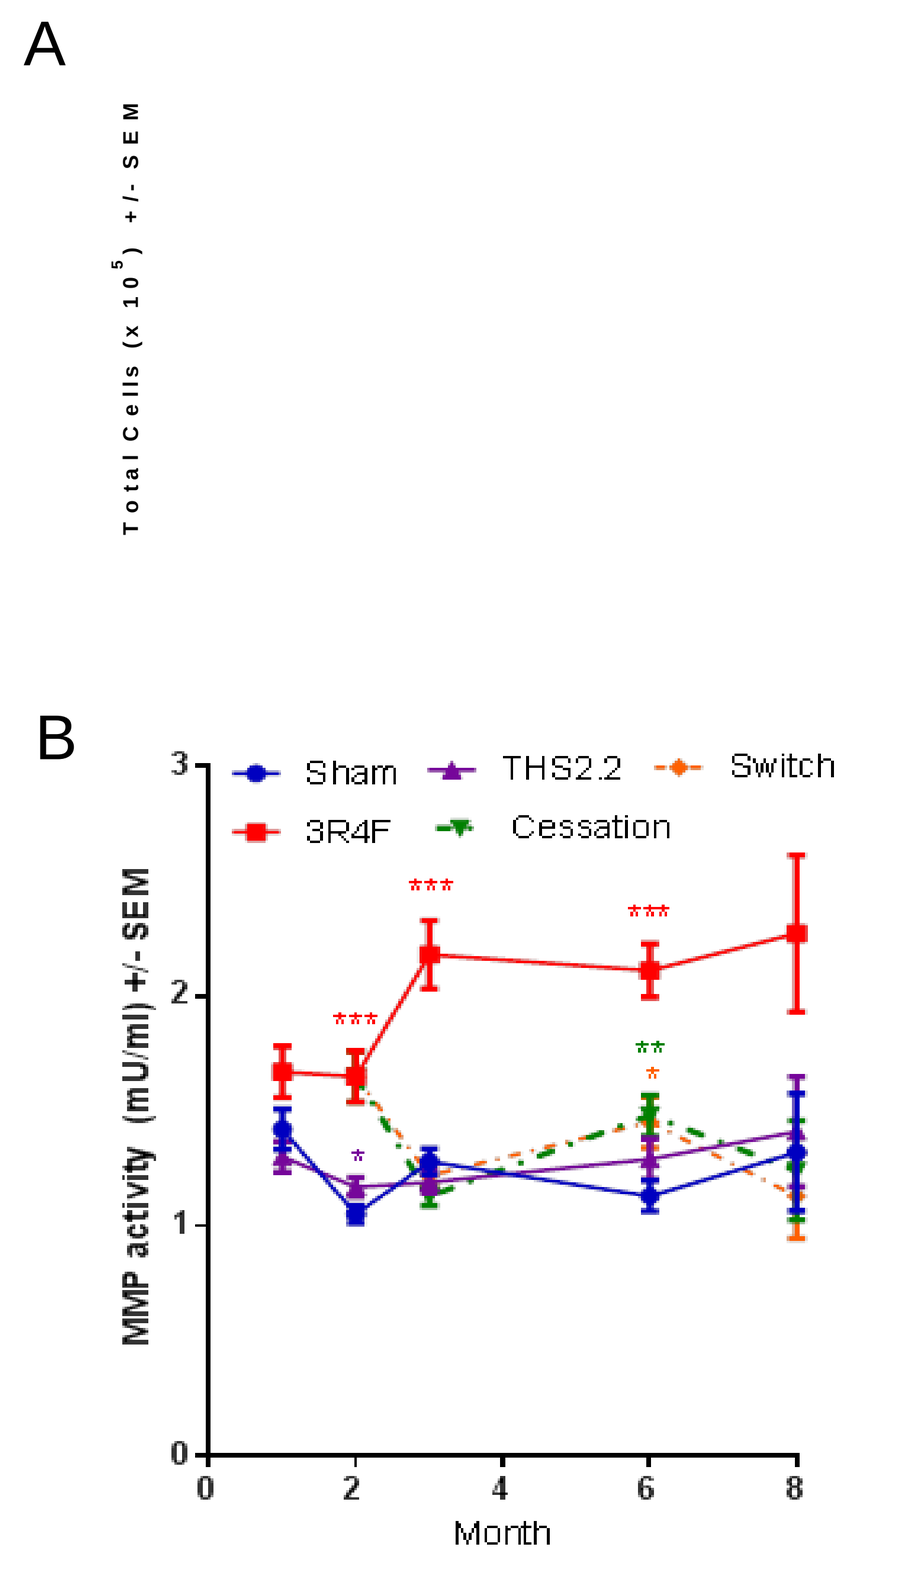
A
Total Cells (x 10^5) +/- SEM
B
Sham
THS2.2
Switch
3R4F
Cessation
MMP activity (mU/ml) +/- SEM
Month

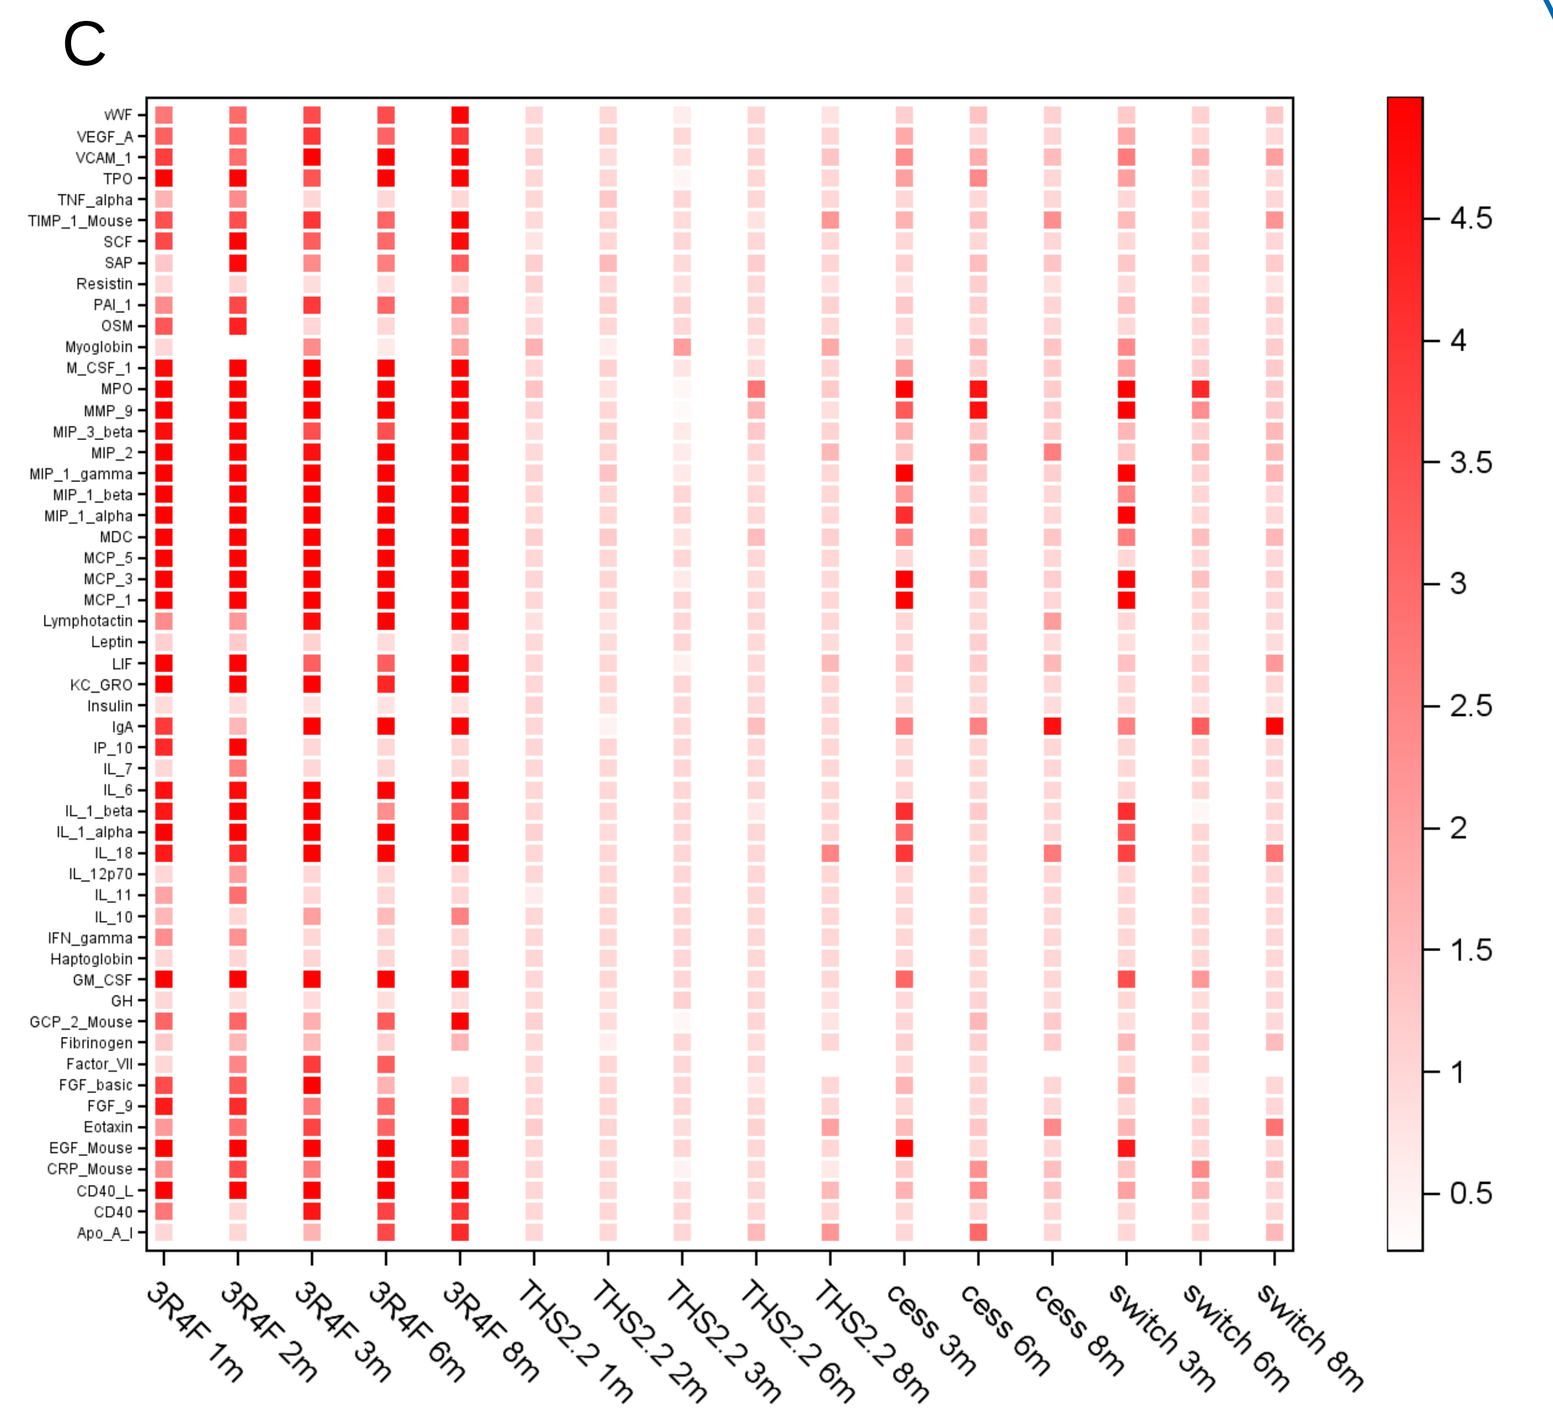
C
Ratio is given by median of treated mice over median of sham mice at same time point (truncated scale)

The absolute number of inflammatory cells in lung as determined by flow cytometry based analysis of free cells in BALF (A), the activity of matrix metalloproteinase (B) and the levels of inflammation-related chemokines detected in bronchoalveolar lavage fluid (C) were dramatically increased even after 1 month of exposure to 3R4F. Most of the inflammatory parameters measured reverted to the levels obtained with continuous exposure to filtered air or THS2.2-exposed animals after the cessation or the switch.

Lung volume and histopathological evaluation

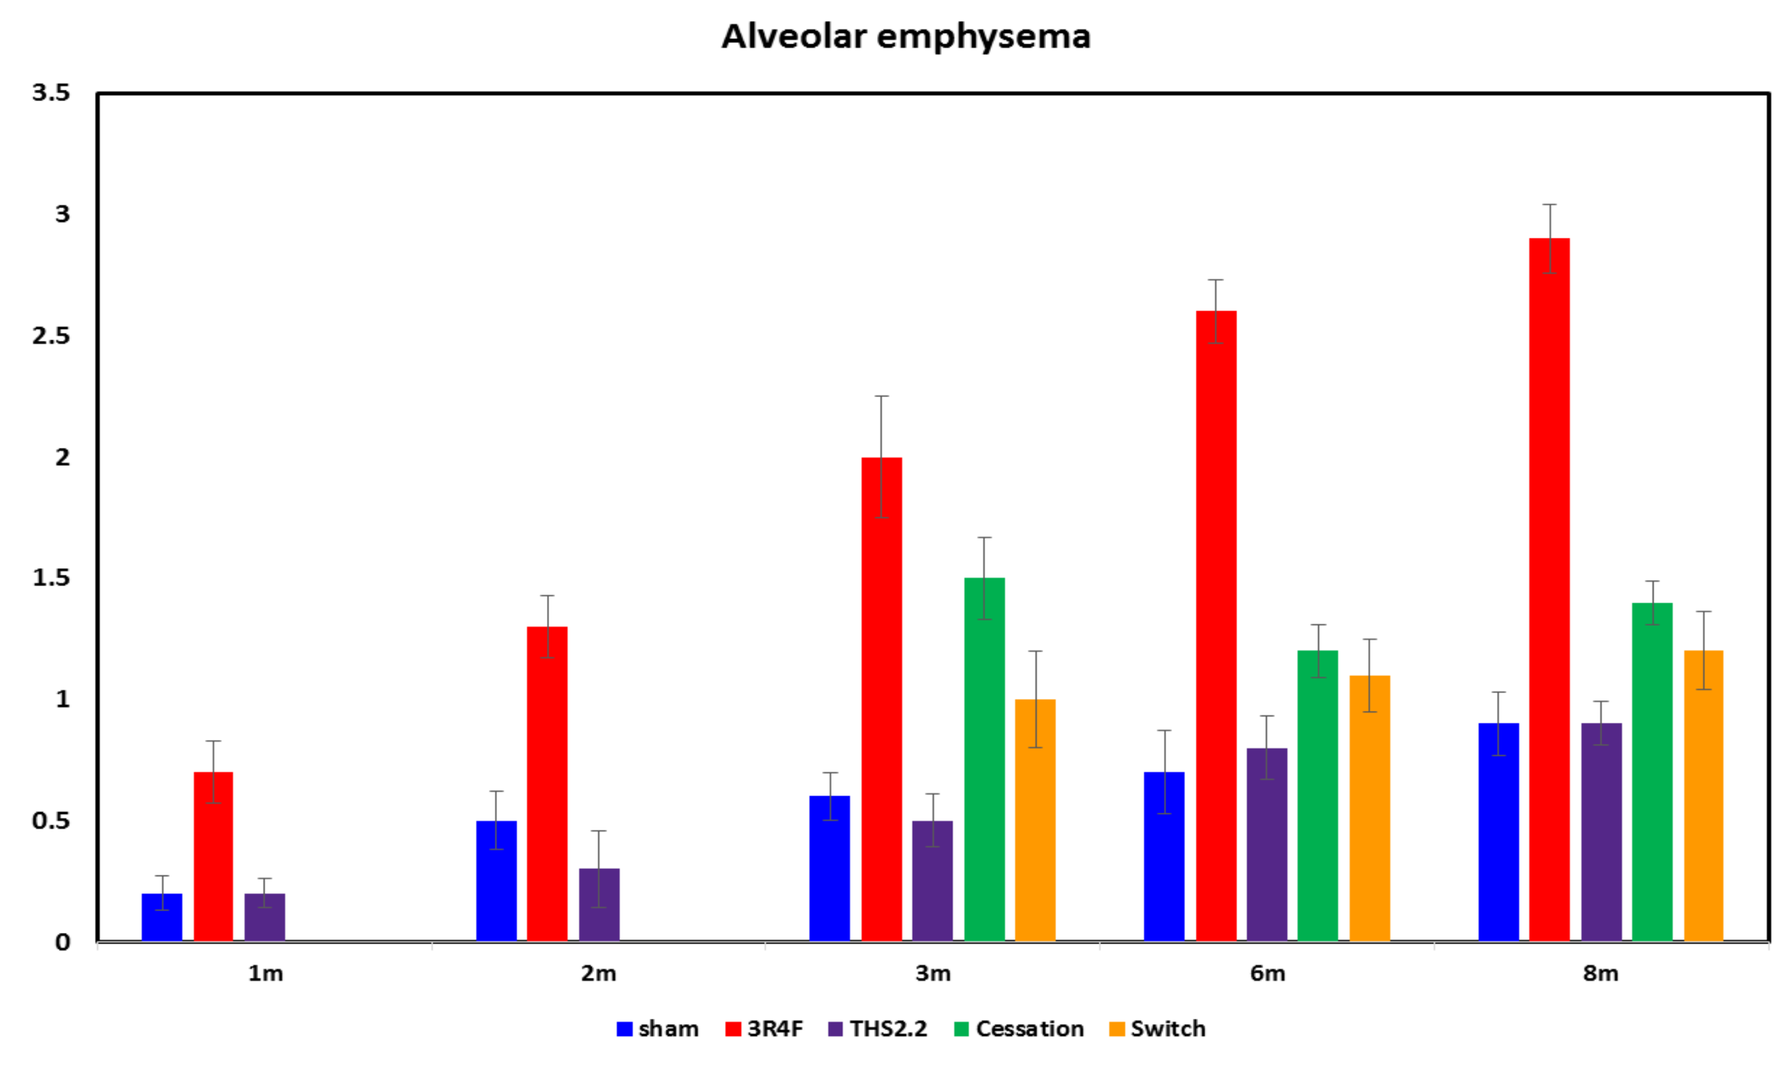
A
Alveolar emphysema
sham
3R4F
THS2.2
Cessation
Switch

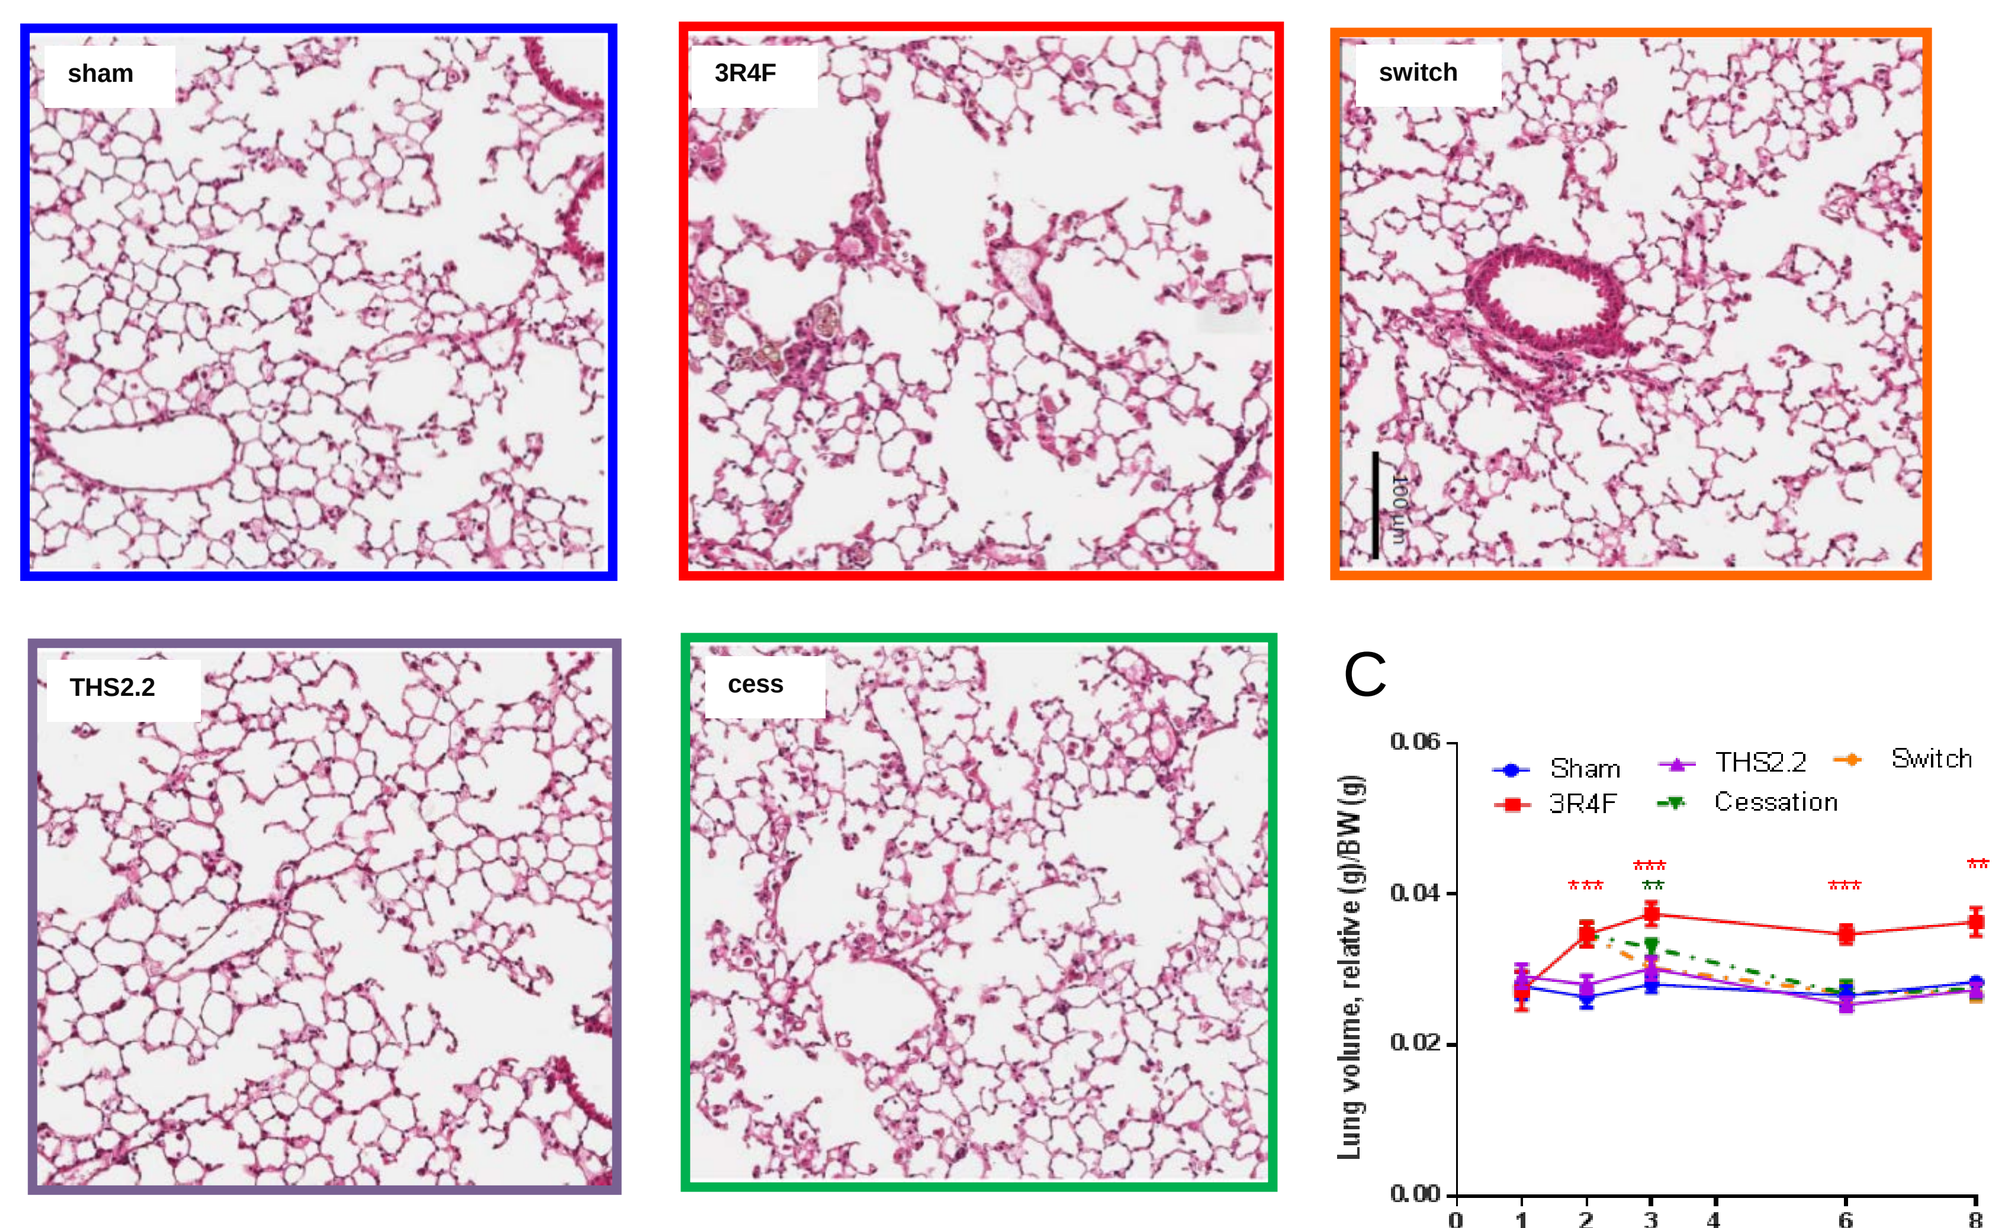
B
sham
3R4F
switch
THS2.2
cess
C
Sham
THS2.2
Switch
3R4F
Cessation
Lung volume, relative (g)/BW (g)
Month

Panel A – Scoring of alveolar emphysema; panel B – Representative haematoxin and eosine stained lung sections; panel C – Lung volume. The level of emphysema was evaluated by the scoring of severity and by morphometrical measurements performed in a blinded manner by an external pathologist. After the switch or cessation, the evaluation revealed a certain level of reduction of severity compared to the emphysema seen in lungs from continuously exposed animals. However, even 6 months after the switch some tissue degradation was still evident (A and B). Those changes are well reflected in the liquid displacement based lung volume measurement (C).

Pulmonary function: pressure-volume (P-V) loops

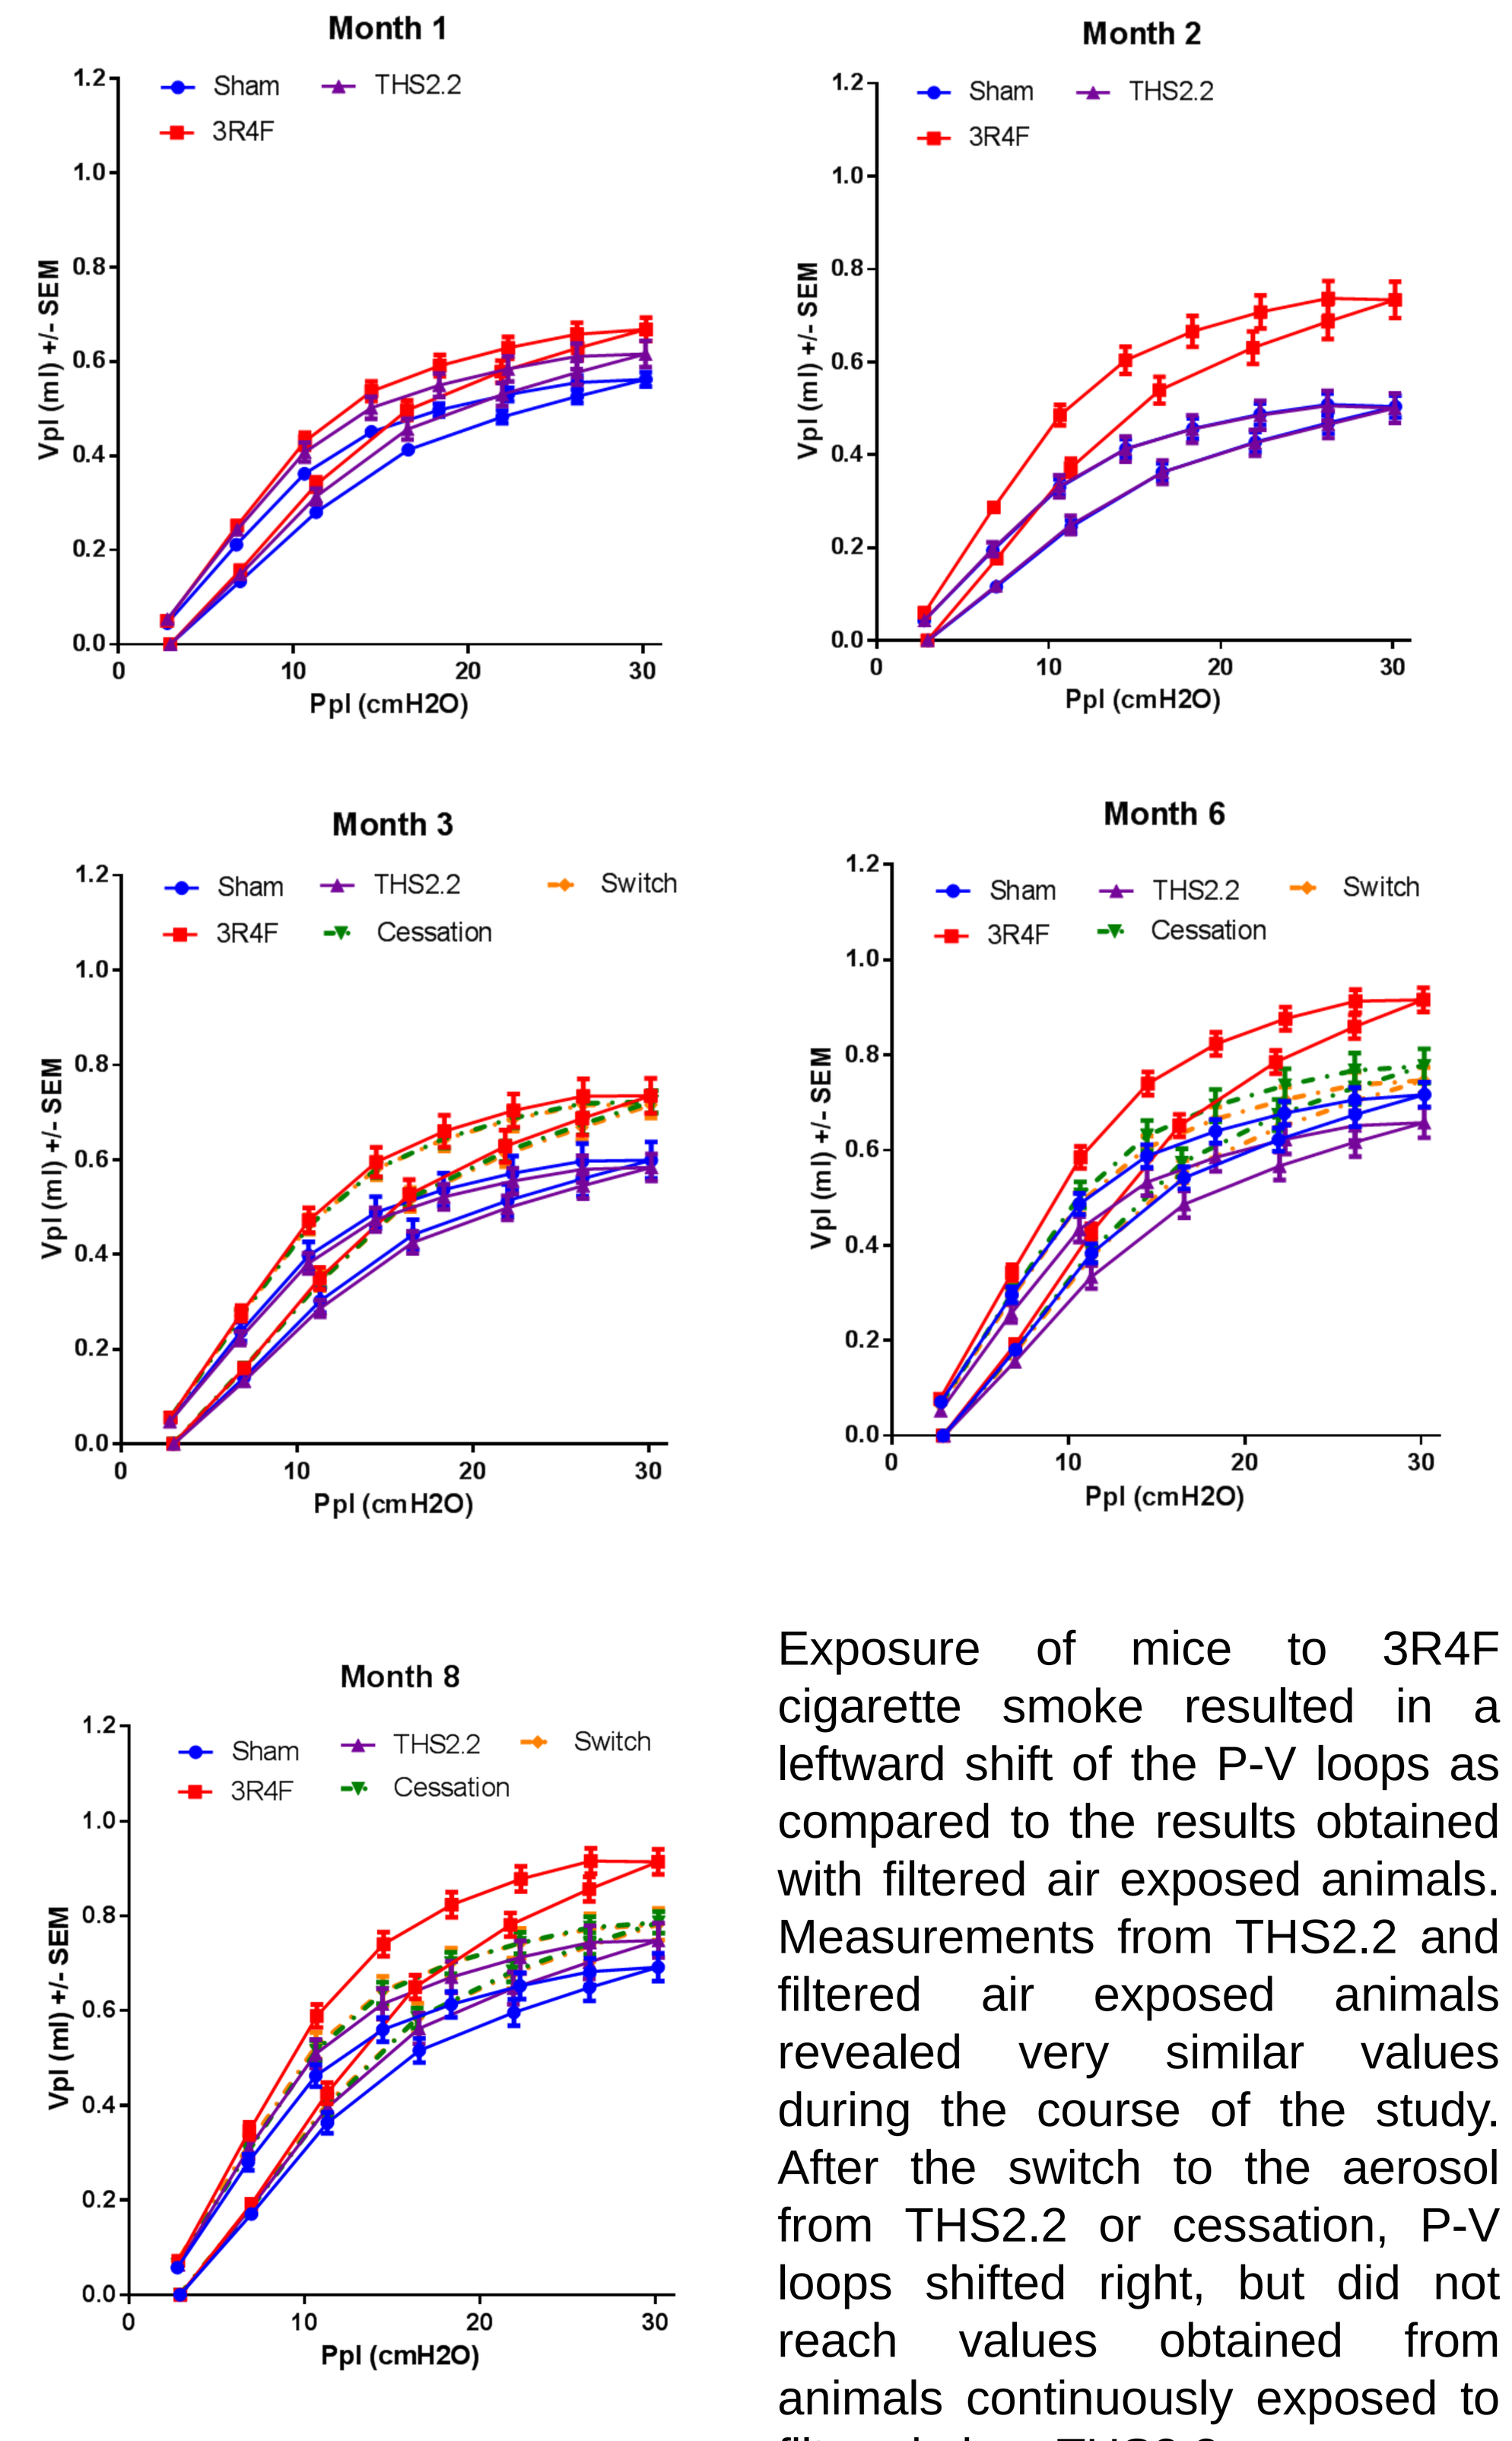
Month 1
Month 2
Month 3
Month 6
Month 8
Sham
THS2.2
3R4F
Switch
Cessation
Vpl (ml) +/- SEM
Ppl (cmH2O)

Exposure of mice to 3R4F cigarette smoke resulted in a leftward shift of the P-V loops as compared to the results obtained with filtered air exposed animals. Measurements from THS2.2 and filtered air exposed animals revealed very similar values during the course of the study. After the switch to the aerosol from THS2.2 or cessation, P-V loops shifted right, but did not reach values obtained from animals continuously exposed to filtered air or THS2.2.

Plaque area measurements

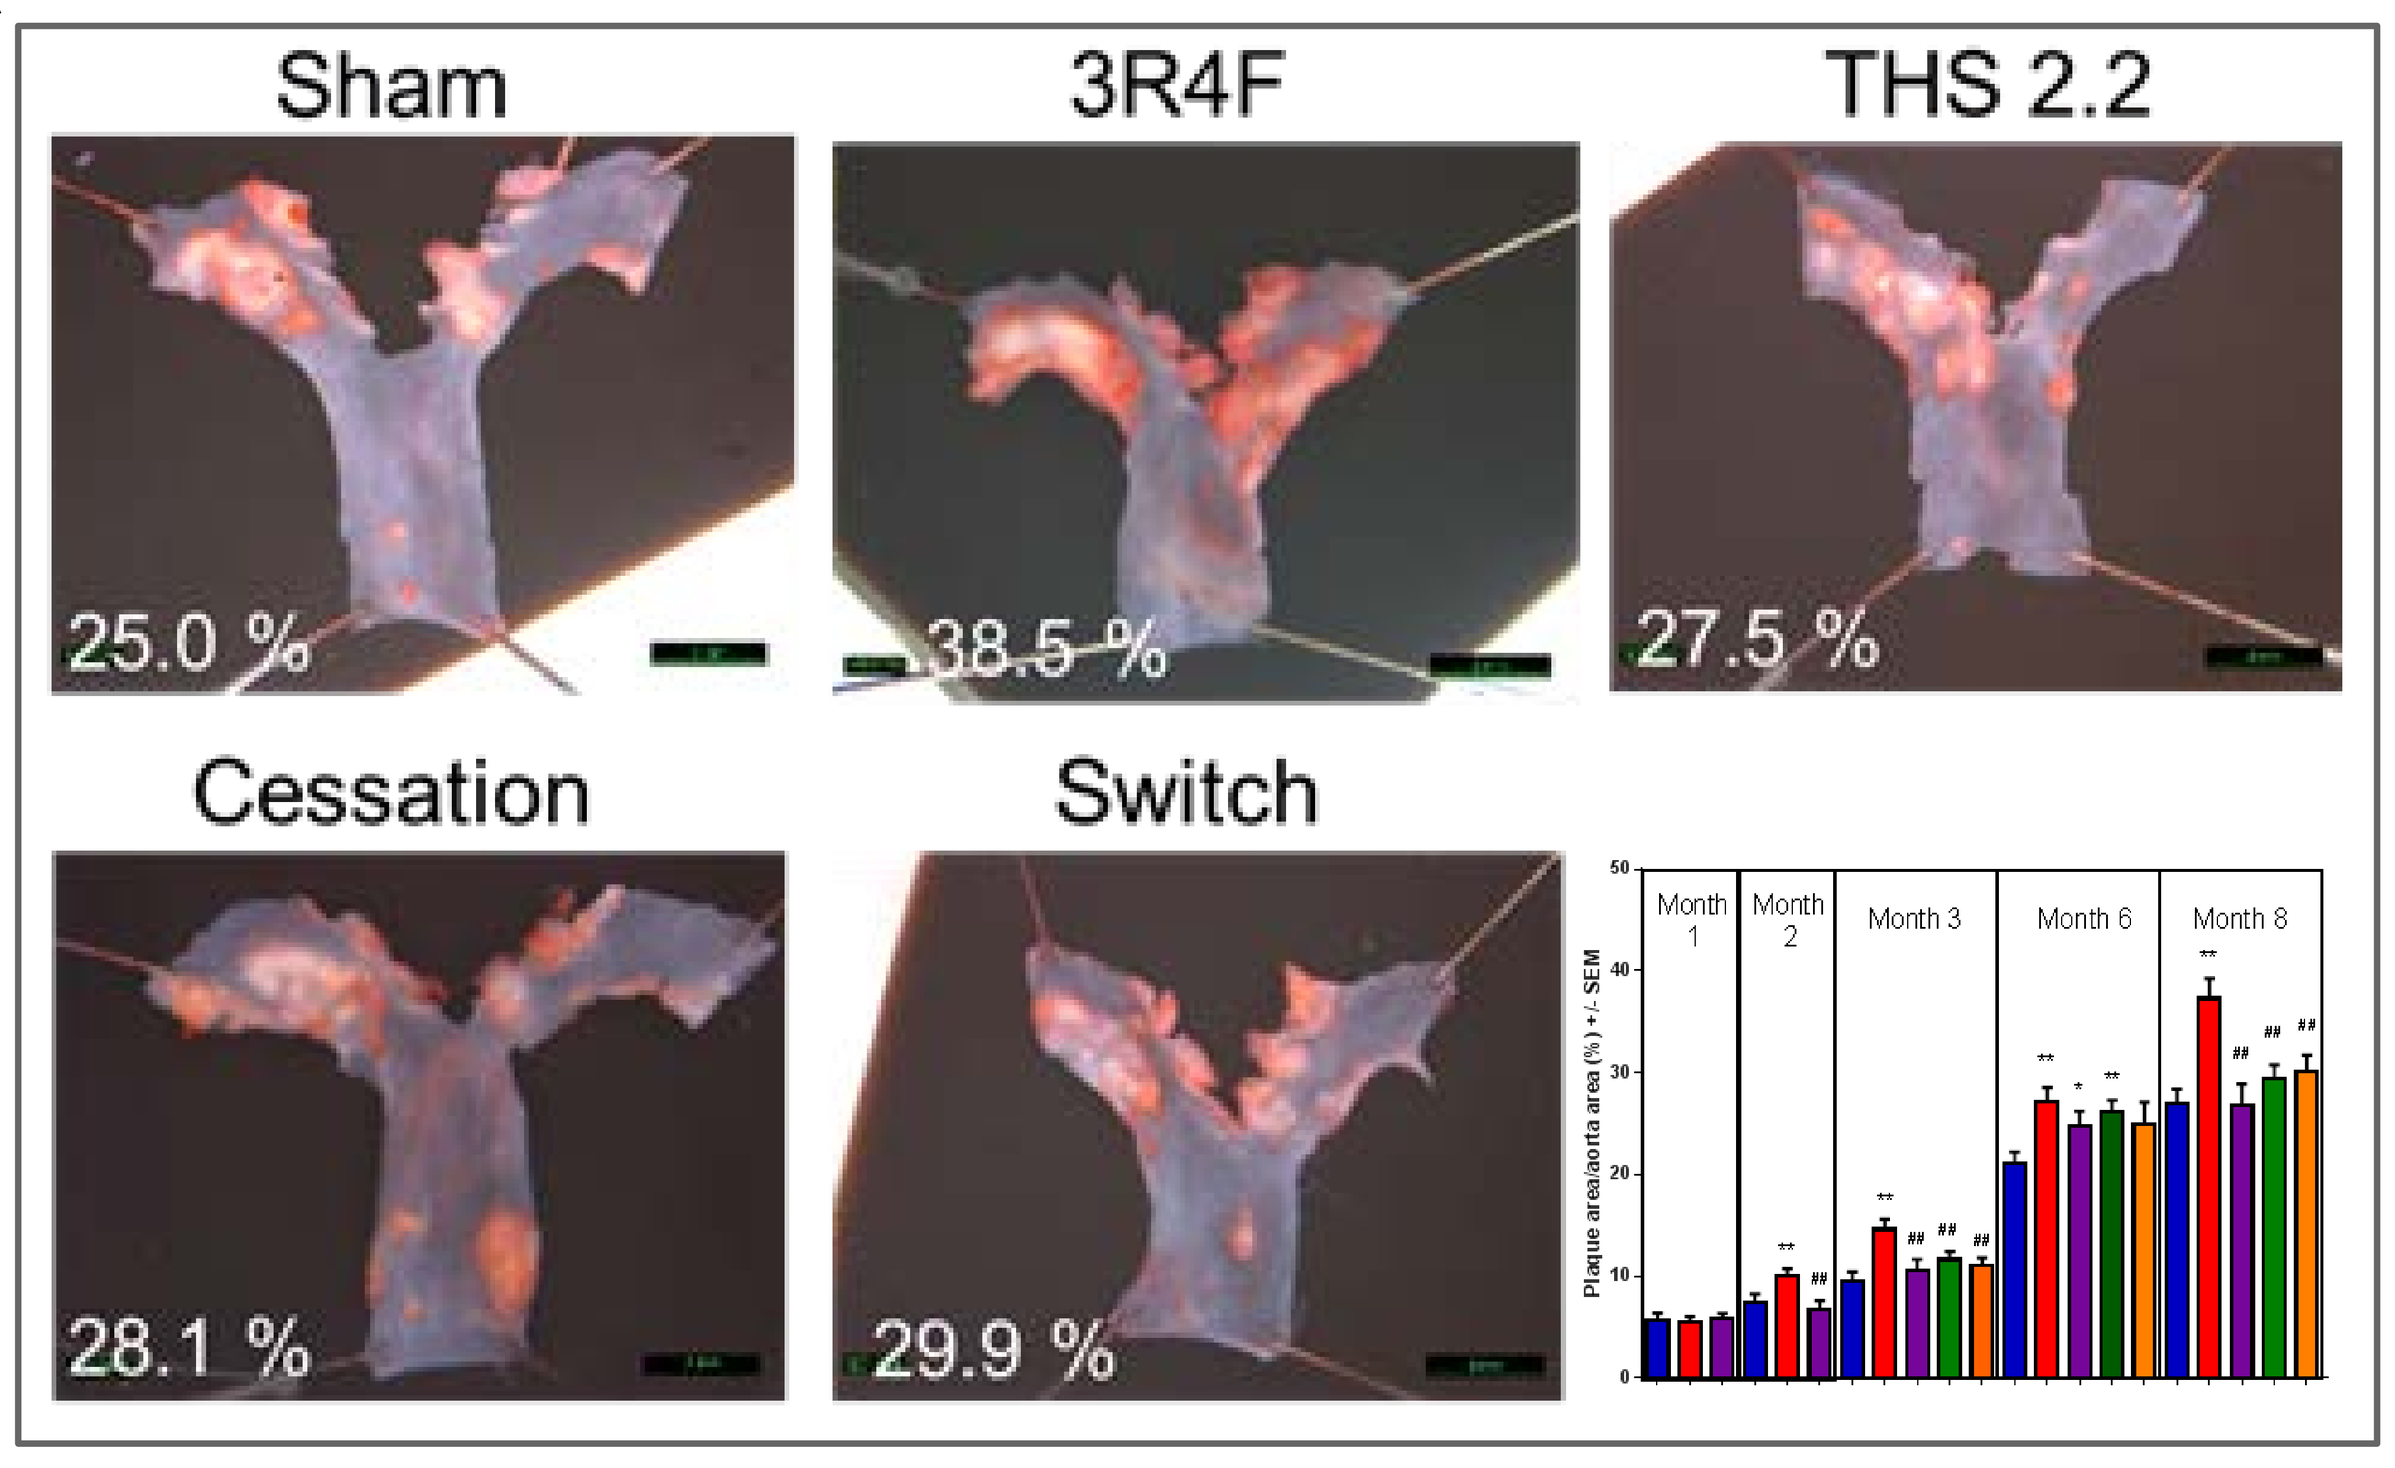
A
Sham
3R4F
THS 2.2
Cessation
Switch
25.0 %
38.5 %
27.5 %
28.1 %
29.9 %
Month 1
Month 2
Month 3
Month 6
Month 8

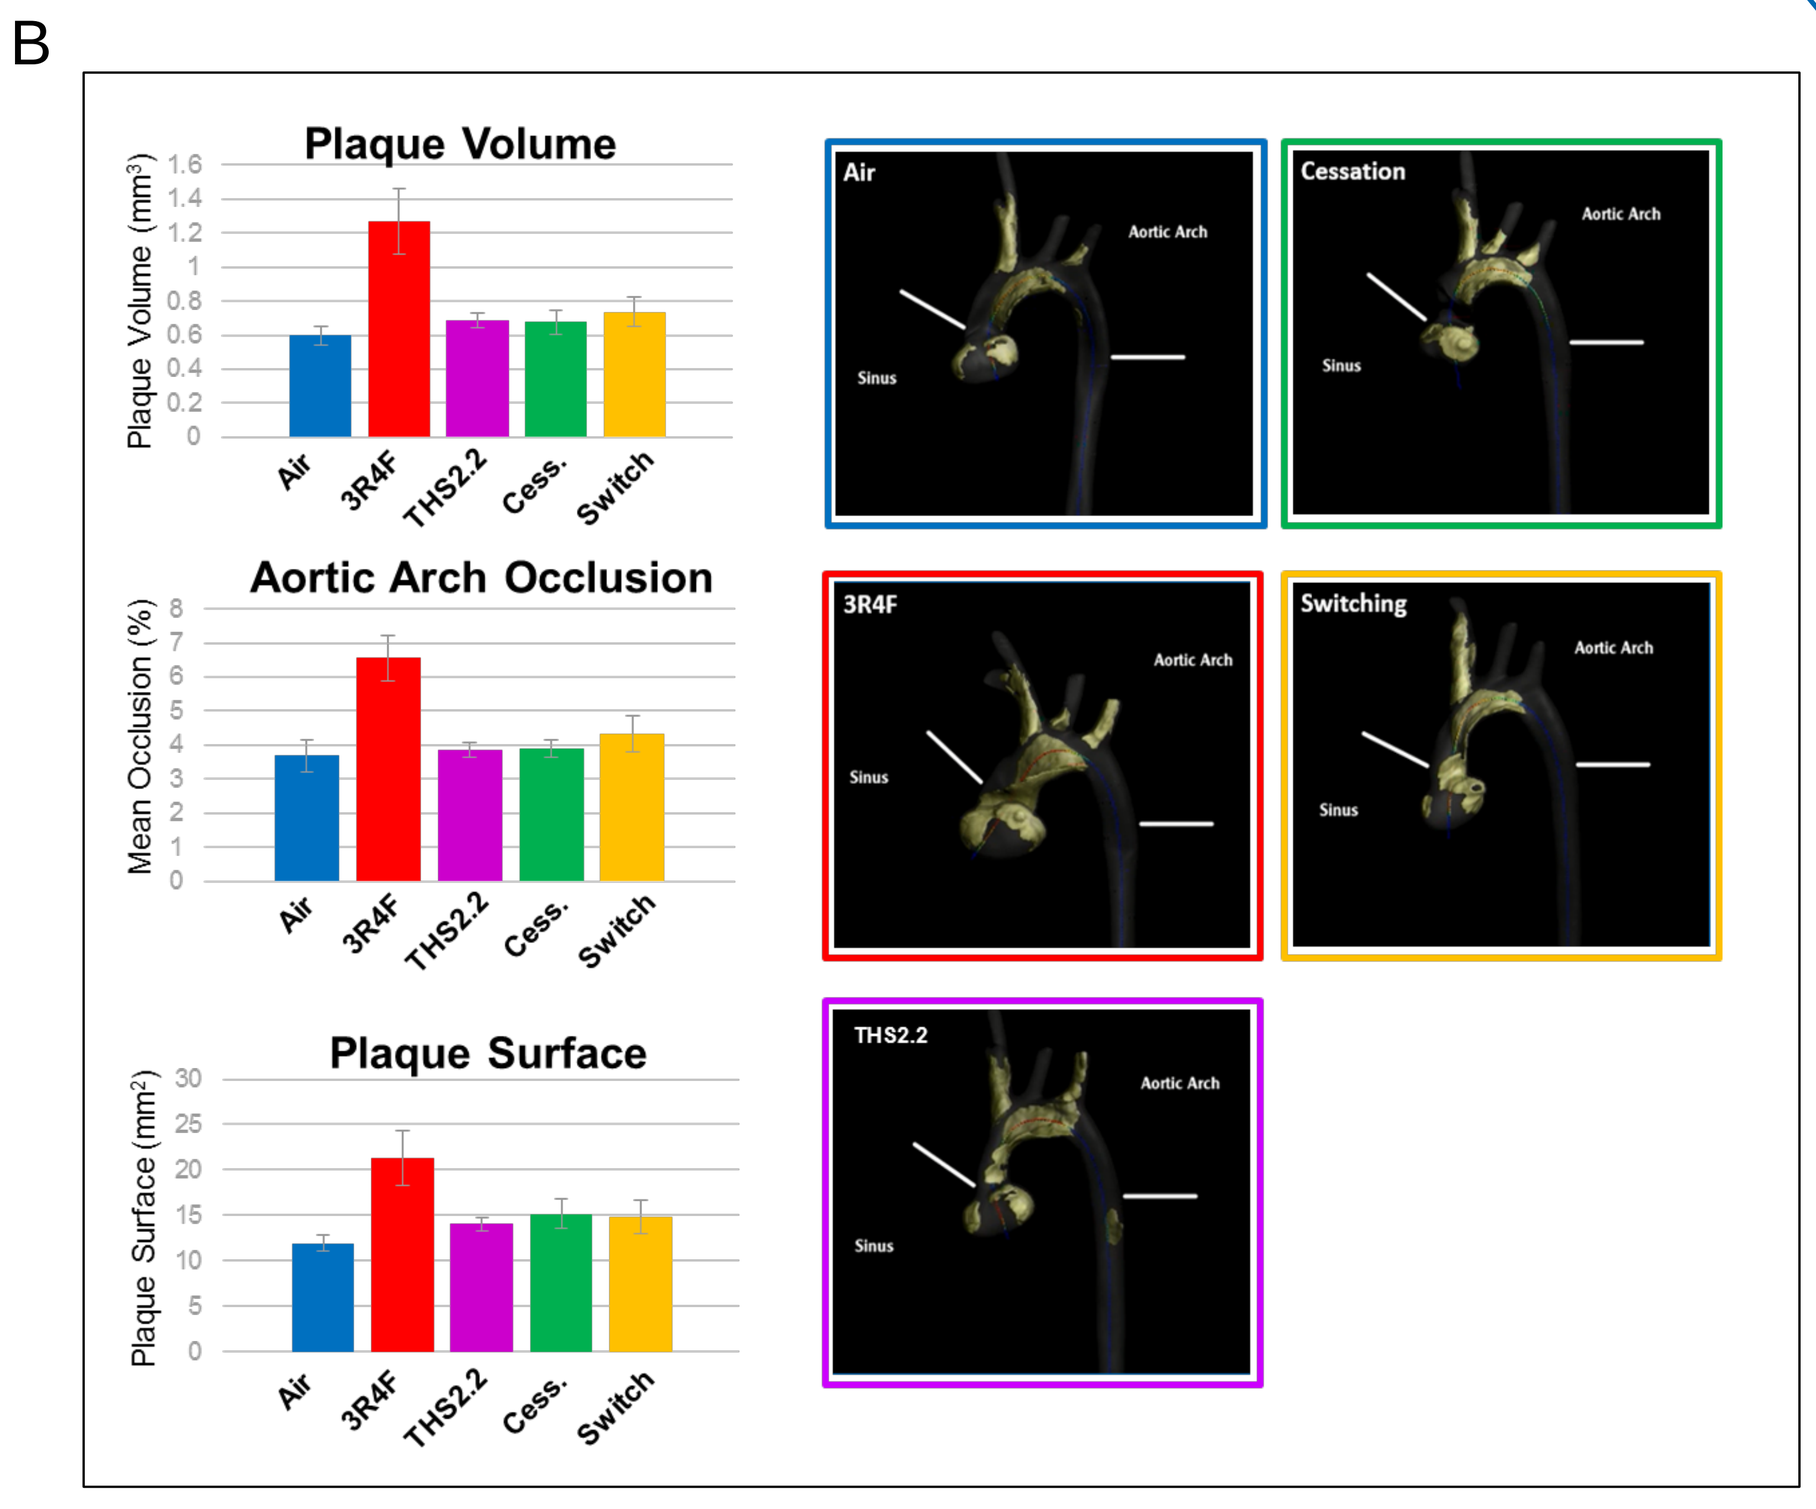
B
Plaque Volume
Aortic Arch Occlusion
Plaque Surface
Air
3R4F
THS2.2
Cess.
Switch
Cessation
Switching
Aortic Arch
Sinus

Exposure to 3R4F resulted in increased plaque formation in aortic arch of ApoE $^{-/-}$ mice compared with sham exposure. These changes were alleviated by smoking cessation or switching to THS2.2 within one month, and nearly reached control levels at the 8-month time point. Panel A – oil red O stained plaque, two-dimensional quantification, percentages indicate the surface of aorta under plaque; panel B – µCT scanned aorta after the staining, 3D reconstruction showing position and the thickness of the plaque (collaboration with an external partner).

Gene expression profiles from lung and nasal epithelium

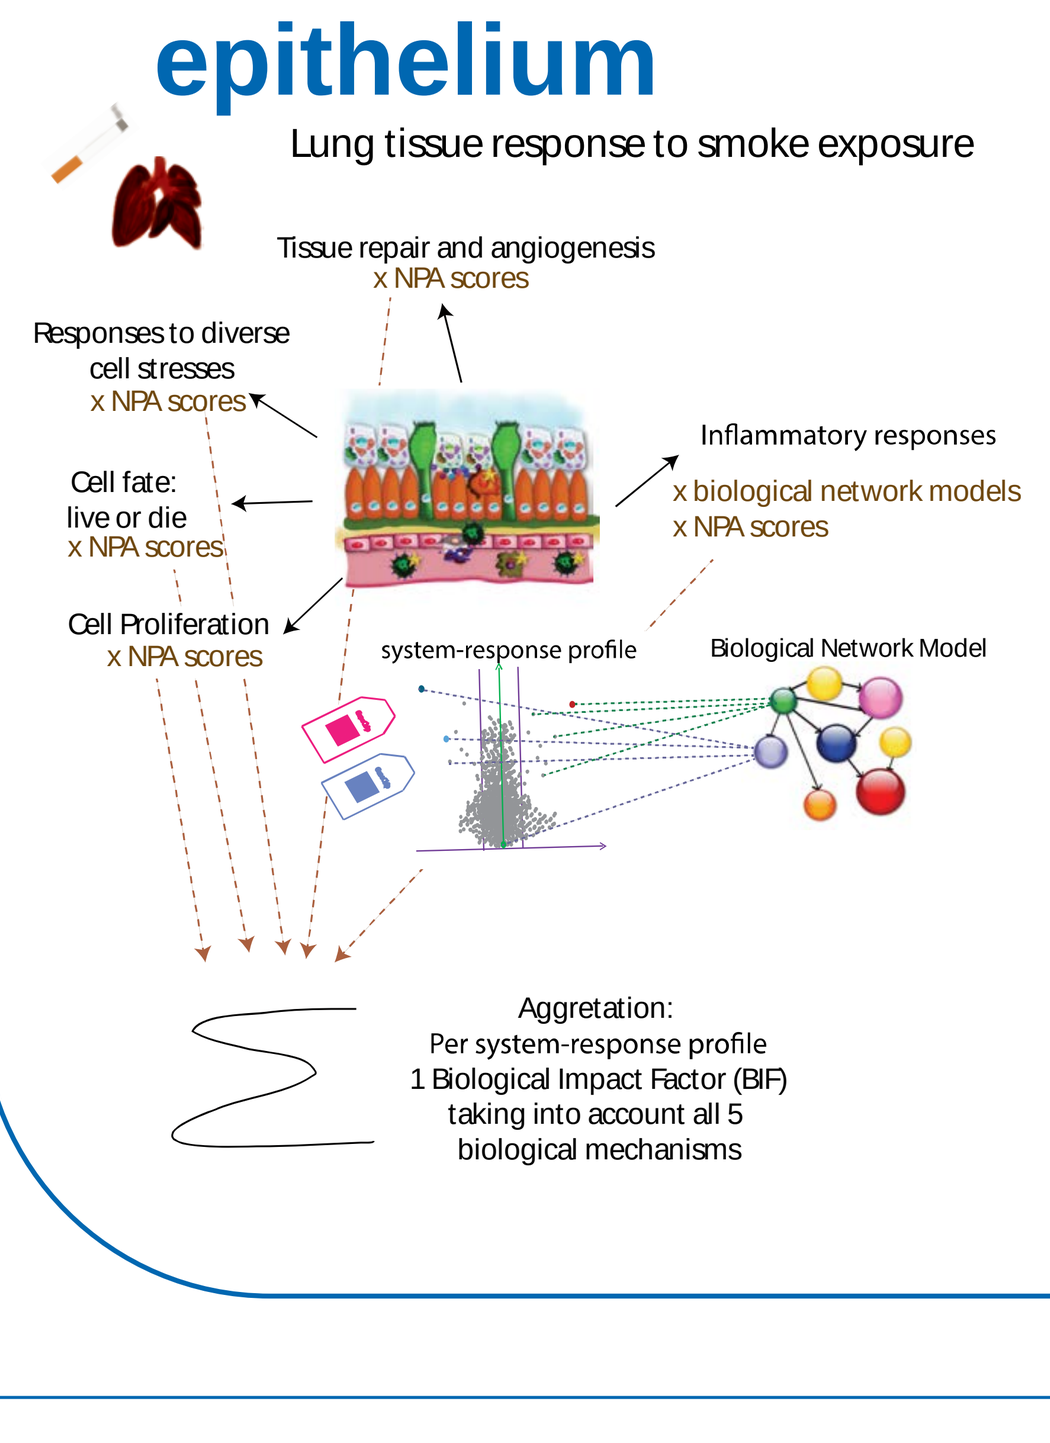
Lung tissue response to smoke exposure
Tissue repair and angiogenesis x NPA scores
Responses to diverse cell stresses x NPA scores
Inflammatory responses x biological network models x NPA scores
Cell fate: live or die x NPA scores
Cell Proliferation x NPA scores
system-response profile
Biological Network Model
Aggretation: Per system-response profile 1 Biological Impact Factor (BIF) taking into account all 5 biological mechanisms

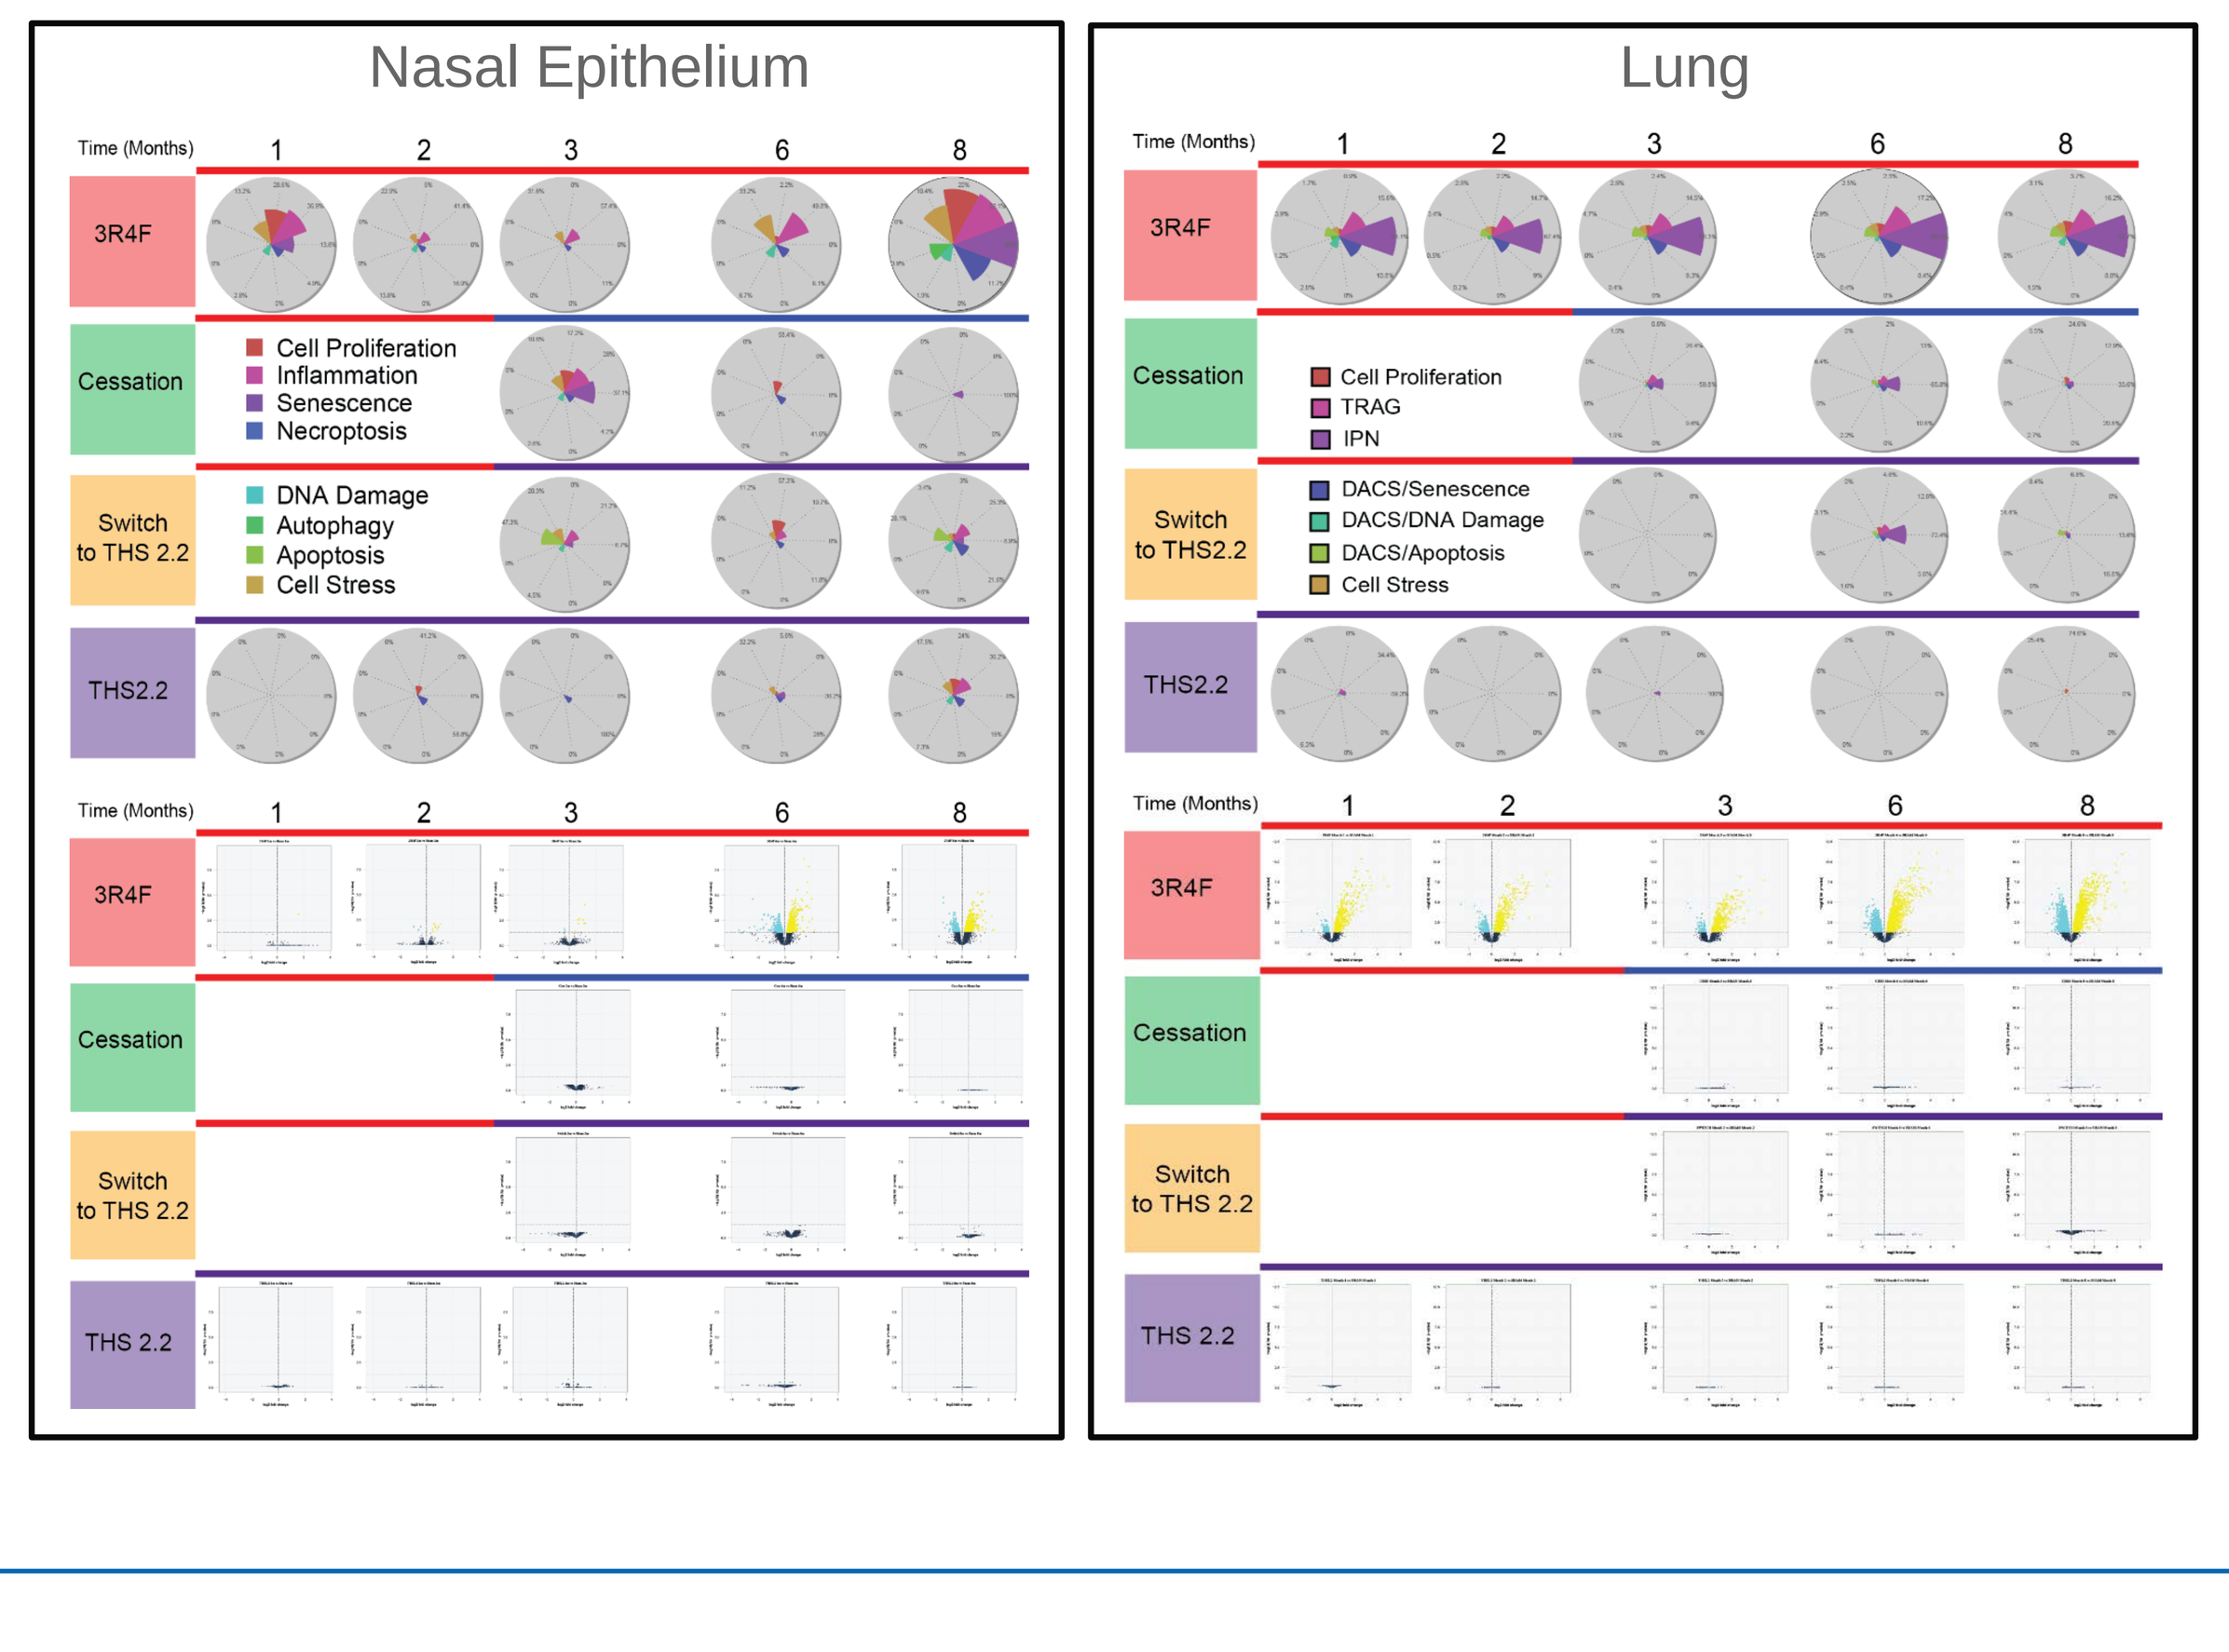
Nasal Epithelium
Lung
Time (Months)
3R4F
Cessation
Switch to THS 2.2
THS2.2
Cell Proliferation
Inflammation
Senescence
Necroptosis
DNA Damage
Autophagy
Apoptosis
Cell Stress
TRAG
IPN
DACS/Senescence
DACS/DNA Damage
DACS/Apoptosis

For each gene, the gene expression change, calculated as the log2 fold change, is plotted on the x-axis and the statistical significance, proportional to the negative log10-adjusted *P*-value, is plotted on the y-axis. Yellow and blue dots highlight genes that are statistically significantly up- or down-regulated, respectively, compared with the sham group at each respective time point. Star plots illustrate the decomposition of the overall transcriptional changes into its eight mechanistic components (from cell proliferation to inflammation, indicated by colours) for each treatment group.

- The exposure to 3R4F cigarette smoke resulted in significant levels of pulmonary inflammation, decline in pulmonary function, and histopathological changes. These phenotypic changes were coherent with the molecular data.
- Chronic exposure to an aerosol from the THS2.2 resulted in very little difference in all measured parameters related to COPD and CVD when compared to the filtered air-exposed animals.
- The biological response to switching to a THS2.2 aerosol or filtered air following 2 months of 3R4F cigarette smoke exposure were very similar between the two conditions across the spectrum of endpoints assessed, and showed a generally positive effect.
- Differential 'omics' profiles associated with 3R4F exposure returned to nearly filtered air-like level following either switching to a THS2.2 aerosol or filtered air.
- Histopathological assessment also showed a marked effect of switching, in which a partial or complete (depending on the inflammatory cell type) reversal of pulmonary inflammation was observed.
- These data collectively indicate a halting or regression of the disease genesis following switching from conventional cigarette to THS2.2 aerosol in ApoE$^{-/-}$.

PMI RESEARCH & DEVELOPMENT

Philip Morris International R&D, Philip Morris Products S.A., Quai Jeanrenaud 5, 2000 Neuchâtel, Switzerland
(Part of Philip Morris International group of companies)