PHARMACOKINETICS OF NICOTINE AND SUBJECTIVE EFFECTS FOLLOWING THE SINGLE USE OF A NON-MENTHOL AND MENTHOL VERSION OF TOBACCO HEATING SYSTEM 2.2 IN TWO STUDIES IN JAPAN: A COMPARISON WITH SINGLE USE OF A COMBUSTIBLE CIGARETTE AND NICOTINE GUM

P. Picavet[1], C. Haziza[1], G. Baker[1], N. Lama[1], J. Ancerewicz[1], M. Benzimra[1], Fumimasa Nobuoka[2], Masayuki Sugimoto[3] and F. Lüdicke[1],

[1] Philip Morris International Research & Development, Neuchâtel, Switzerland; [2] Ageo Medical Clinic, Haraichi, Ageo City Saitama, JP, [3] Koganeibashi Sakura Clinic, Koganei-shi, Tokyo, JP

Introduction and Objectives

Philip Morris International is currently developing potentially reduced risk products (RRPs) with the intention to reduce the risk of tobacco-related diseases. The challenge in developing RRPs is two-fold, i.e., developing tobacco products that are shown to reduce risk and that are acceptable to smokers as substitutes for combustible cigarettes (CC). The candidate RRP, the Tobacco Heating System (THS) 2.2, tested in this study is heated at significantly lower temperatures than required for CC.

The study reported here is part of a global clinical program and the objective of the study was to evaluate the plasma pharmacokinetic (PK) profile of nicotine following single use of THS 2.2 menthol and non-menthol as compared to menthol and non-menthol combustible cigarettes (CC) and nicotine replacement therapy (NRT), respectively. Subjective effects were evaluated to get first insight to which extent adult smokers would find THS 2.2 an acceptable substitute for CC.

Methods

The two studies were open-label, randomized, two-period, four-sequence crossover studies in 62 healthy smokers. Each period consisted of 2 days, with 1 day of smoking abstinence (nicotine wash-out) and 1 day of single use THS 2.2, CC or nicotine gum with every subject being exposed to 2 of the 3 study products (THS 2.2/CC and THS 2.2/nicotine gum [NRT]). During the single use day, a total of 16 venous blood samples were collected including 1 sample prior to product use and at various time points for up to 24 hours.

One study (Study PK-02) tested the THS 2.2 non-menthol (THS 2.2), the second study (Study PK-05) tested the THS 2.2 menthol (mTHS 2.2) product. The International Organization on Standardization (ISO) yield per THS 2.2 was 0.5 mg nicotine. The (ISO) yield for mTHS 2.2 was 0.5 mg nicotine.

Nicotine concentration was determined in plasma using a validated method (LC-MS/MS; LLOQ: 0.2 mg/ml). Urge to smoke was assessed using the questionnaire of smoking urges-brief (Cox et al., 2001).

The studies were registered with ClinicalTrials.gov (NCT01959607/NCT01967706). The studies were approved by Institutional Review Board and were conducted in Tokyo, Japan in 2013 in accordance with ICH GCP guidelines.

Results

Demographics

Summary of Demographics and Baseline Characteristics

		Study PK-02 (n=62)	Study PK-05 (n=62)
Sex (male) [n (%)]		34 (55)	32 (52)
Age [yr M ± SD]		33.6 ± 9.2	32.8 ± 9.5
Body Mass Index [kg/m² M ± SD]		22.9 ± 3.1	22.5 ± 2.4
Nicotine ISO Yield [n (%)]	≤ 0.6 mg	32 (52)	36 (58)
	> 0.6 - 1 mg	30 (48)	26 (42)
Number of CC/Day [n (%)]	10 - 19	34 (55)	36 (58)
	> 19	28 (45)	26 (42)

Nicotine PK Endpoints Parameters

The overall shape of the nicotine concentration-time curves for THS and CC were similar in both studies and differed for THS and NRT gum. The values for $AUC_{(0\text{-last})}$ and C_{max} were comparable for THS and CC in both studies. The t_{max} was similar for CC and THS (about 6 min) in both studies.

Primary PK Parameters THS 2.2 vs CC

PK Parameter (unit)	Product Exposure	N	Geometric Means	THS 2.2/CC Ratio	95% CI (%)
$AUC_{(0\text{-last})}$ (ng.h/mL)	THS 2.2	42	23.8	96.3	(85.1-109.1)
	CC	42	24.7		
C_{max} (ng/mL)	THS 2.2	42	14.3	103.5	(84.9-126.1)
	CC	42	13.8		

Primary PK Parameters mTHS 2.2 vs mCC

PK Parameter (unit)	Product Exposure	N	Geometric Means	mTHS 2.2/mCC Ratio	95% CI (%)
$AUC_{(0\text{-last})}$ (ng.h/mL)	mTHS 2.2	43	24.0	98.1	(80.6-119.5)
	mCC	43	24.5		
C_{max} (ng/mL)	mTHS 2.2	43	10.7	88.5	(68.6-114.0)
	CC	43	12.1		

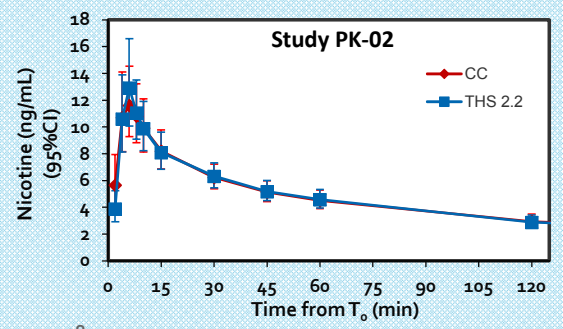
Study PK-02
CC
THS 2.2
Nicotine (ng/mL) (95%CI)
Time from T_o (min)

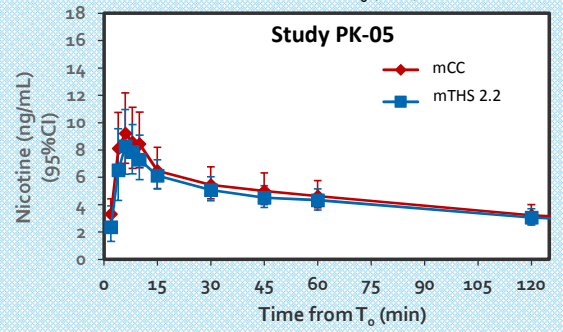
Study PK-05
mCC
mTHS 2.2
Nicotine (ng/mL) (95%CI)
Time from T_o (min)

In Study PK 02 the C_{max} (11.5 ng/mL for THS 2.2; 4.8 ng/mL for NRT) and $AUC_{(0\text{-last})}$ (18.9 ng*h/mL for THS 2.2; 14.88 ng*h/mL for NRT) were higher for THS 2.2 compared to NRT, whereas the C_{max} (7.6 ng/mL for mTHS 2.2; 7.5 ng/mL for NRT) was comparable for mTHS 2.2 and NRT in Study PK 05 with the $AUC_{(0\text{-last})}$ (15.6 ng*h/mL for mTHS 2.2; 27.9 ng*h/mL for NRT) for mTHS 2.2 being lower than NRT.

Urge to Smoke Symptoms (QSU-Brief) Following Single Use

For THS and CC there was an approximately 35% reduction in the average urge to smoke total score observed 15–30 minutes after single use in both studies. Overall, the average QSU-Brief total score was similar for THS compared to CC following single use. Compared to NRT gum use, the mean total QSU-Brief score over time points was lower following THS use.

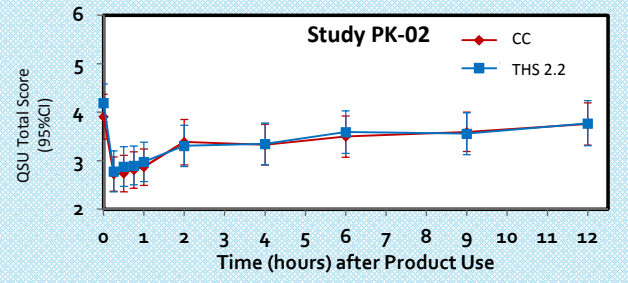
Study PK-02
CC
THS 2.2
QSU Total Score (95%CI)
Time (hours) after Product Use

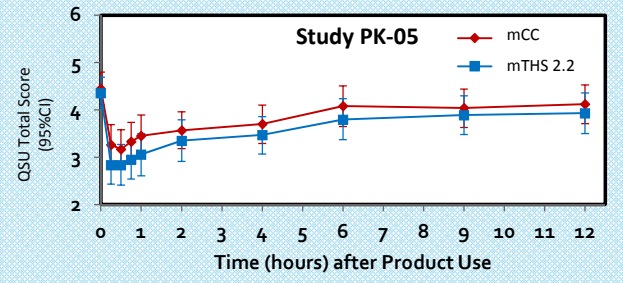
Study PK-05
mCC
mTHS 2.2
QSU Total Score (95%CI)
Time (hours) after Product Use

QSU-Brief Results THS 2.2 vs CC

Study	Product	Baseline	Minimum	% Change from Baseline	Timepoint
PK-02	THS 2.2	4.2	2.8	-34%	15min
	CC	3.9	2.7	-30%	15min
PK-05	mTHS 2.2	4.4	2.8	-35%	15min
	mCC	4.5	3.2	-29%	30min

Safety

No serious or severe adverse events (AEs) were reported in the studies. The pooled incidence and frequency of AEs were low with 18 AEs reported in 15 subjects after randomization. Three AEs were related to the THS 2.2 or CC, and 5 AEs were related to study procedures. No AEs were related to NRT gum. The most frequent AEs were hemoglobin decreased, bilirubin increased, blood triglycerides increased, and dysphoria.

Conclusions

The PK profiles for both THS 2.2 variants evaluated were comparable to CC and different from nicotine gum in both studies. A transient reduction in urge-to-smoke was observed with THS 2.2, comparable to CC and higher than nicotine gum after single use.

REFERENCES

Cox LS, Tiffany ST, Christen AG. Evaluation of the brief questionnaire of smoking urges (QSU-brief) in laboratory and clinical settings. Nicotine Tob Res. 2001;3:7-16.

PMI SCIENCE
PHILIP MORRIS INTERNATIONAL

SRNT – 21ST Annual Meeting, Philadelphia, USA
25-28 February 2015

COMPETING FINANCIAL INTERESTS

The research described in this poster was sponsored by Philip Morris International