Plasma Protein Biomarker Identification for Monitoring Molecular Changes in Smokers, Former Smokers, and E-Vapor Product Users Compared with Never Smokers

Catherine Nury[1], Yvan Eb-Levadoux[1], Aditya Kolli[1], Carine Poussin[1], Timothy M. Curran[2], Laurel A. Baglia[2], Rachel A. Berg[2], Loyse Felber Medlin[1], Sam Ansari[1], Edina Kishazi[1], Florian Martin[1], Björn Titz[1], Patrick Picavet[1], Nikolai V. Ivanov[1], Sherry L. Spinelli[2], Julia Hoeng[1] and Manuel C. Peitsch[1]

[1]PMI R&D, Philip Morris Products S.A., Quai Jeanrenaud 5, CH-2000 Neuchâtel, Switzerland
[2]University of Rochester Medical Center, Rochester, New York, USA

Introduction

Inhaled toxicants present in tobacco smoke accelerate and exacerbate inflammation and oxidative stress. Over time, these processes increase the risk of developing respiratory and cardiovascular diseases (CVD). Developing alternative tobacco or nicotine products that reduce health risks for smokers is one approach for addressing these health concerns. The electronic (e)-vapor product (eCig) is proposed as a potential reduced-risk product; however, its health impact requires in-depth investigation. The goal of this human research study is to examine the molecular profiles and biological networks that are perturbed by smoking, reversible upon smoking cessation, and largely unknown with respect to EVP usage for identification of new biomarkers.
Here, we present the plasma protein profiling data of this study. A total of 205 plasma samples were obtained from smokers (CS), former smokers (FS), and eCig users and compared with those of never smokers (NS).

Study overview

In this study, 205 eligible subjects were enrolled, including 52 NS, 77 CS, 37 FS, and 39 eCig users.

Table 1. Demographics by study group

Variables and statistics	NS	CS	FS	eCig
Total recruitment numbers (eligible)	52	77	37	39
Age, years (mean [SD])	30.1 [7.3]	36.7 [7.4]	34.2 [8.8]	29.0 [8.8]
Sex, %				
- Female	65.4	50.6	64.6	23.1
- Male	34.6	49.4	29.7	76.9
- Other	0	0	2.7	0
Body mass index (BMI), kg/m^2 (mean [SD])	25.6 [4.6]	28.6 [5.8]	29.8 [5.7]	26.4 [5.8]
Waist circumference, cm (mean[SD])	80.5 [11.9]	89.4 [13.7]	94.0 [15.0]	84.8 [10.9]
Ethnicity and race, %				
- Caucasian	42	47	30	26
- Non-Caucasian	10	30	7	13
African American	3	21	3	4
Asian	4	1	1	1
Hispanic	0	3	2	3
Other	3	5	1	5

Acrylonitrile is generated at temperatures ranging from 500°C to 800°C, which makes the 2-cyanoethylmercapturic acid (CEMA) urinary metabolite, a biomarker of exposure to acrylonitrile, a good candidate as a biomarker of verification for subject-reported product use for distinguishing cigarette (CC) smoking from non-combustible tobacco product use.[1] On the basis of CEMA measurements, a "compliant" set of 150 subjects were identified, including 50 NS, 57 CS, 25 FS, and 18 eCig users.

Figure 1. CEMA as "compliance" maker. *A CEMA level of 40 ng/mg of creatinine (red line) was selected to identify the "compliant" set of subjects.*

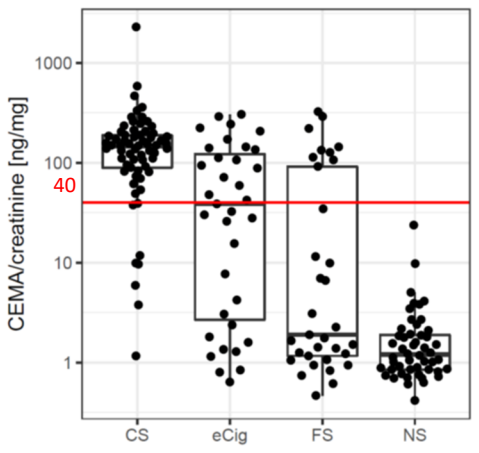
CEMA/creatinine [ng/mg]
1000
100
40
10
1
CS
eCig
FS
NS

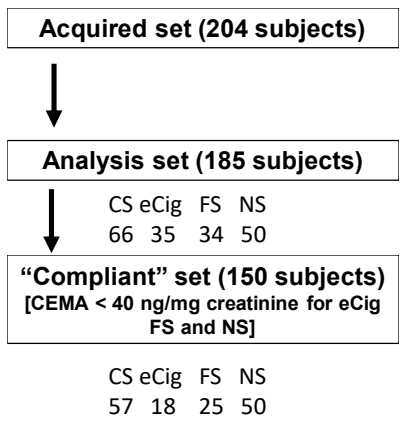
Acquired set (204 subjects)
Analysis set (185 subjects)
CS eCig FS NS
66 35 34 50
"Compliant" set (150 subjects)
[CEMA < 40 ng/mg creatinine for eCig FS and NS]
CS eCig FS NS
57 18 25 50

[1] Minet E, Cheung F, Errington G, Sterz K, Scherer G. *Biomarkers*. 2011;16(1):89-96. doi:10.3109/1354750X.2010.533287

Methods

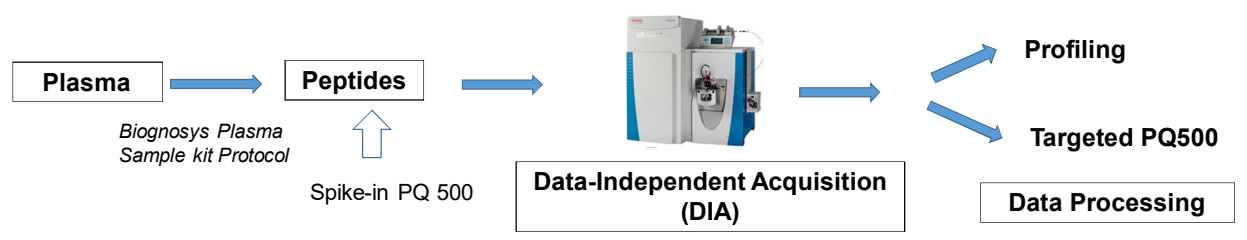
Plasma
Peptides
Biognosys Plasma Sample kit Protocol
Spike-in PQ 500
Data-Independent Acquisition (DIA)
Profiling
Targeted PQ500
Data Processing

Figure 2. Schematic overview of the protocol used for the data-independent acquisition analysis.
Human plasma samples (10 µL) were processed with Sample Preparation Kit Pro from Biognosys in accordance with the manufacturer's protocol. The dried peptide pellets were resuspended in 18 µl LC Buffer and mixed with 6 µL of freshly prepared PQ500 reference peptide stock solution. Peptides were separated on a 15-cm x 1-mm (1.7 µm) C18-CSH column by using a 37-min gradient on a Shimadzu Nexera 2 UPLC connected to a Thermo Qexactive High Field (HF) in data-independent acquisition mode (23 windows). The DIA data were analyzed by using the Direct DIA functionality in Spectronaut version 14.

Targeted analysis (PQ 500) results

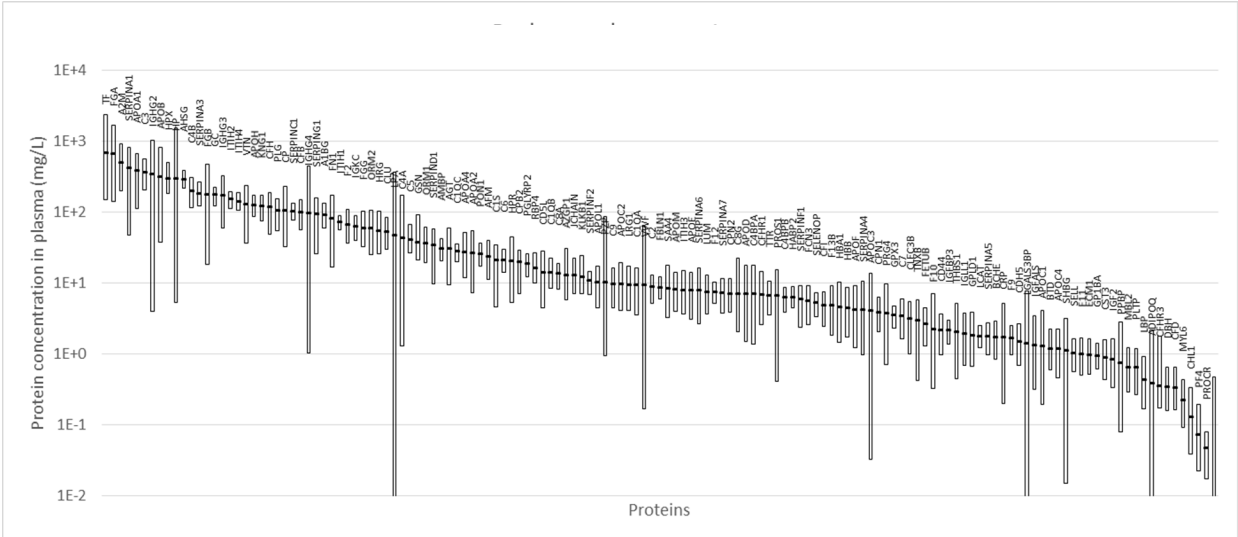
Protein concentration in plasma (mg/L)
1E+4
1E+3
1E+2
1E+1
1E+0
1E-1
1E-2
Proteins

Figure 3. Bar plot of the average concentrations (mg/L) of proteins in plasma on the basis of PQ500 heavy labelled peptides.

On average, robust quantification of 125 protein groups in each of the 204 samples.

Computational analysis of the profiling (DIA) protein set

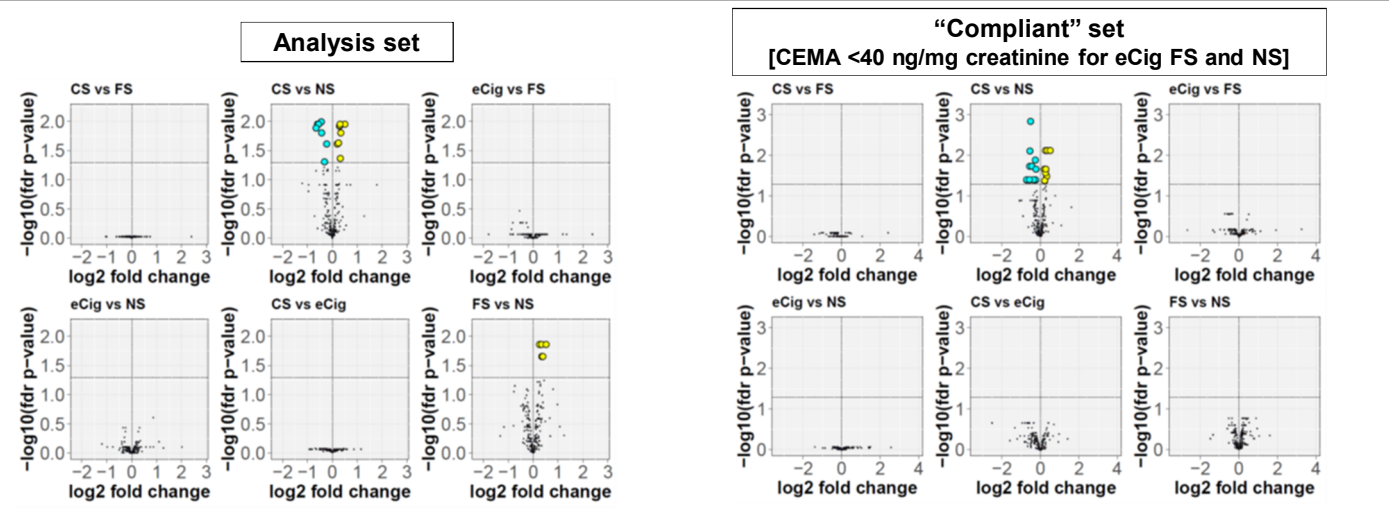
Analysis set
"Compliant" set
[CEMA <40 ng/mg creatinine for eCig FS and NS]
CS vs FS
CS vs NS
eCig vs FS
eCig vs NS
CS vs eCig
FS vs NS
−log10(fdr p−value)
log2 fold change

Figure 4. Volcano plots representing the system response profiles. *For each protein, the protein expression change, calculated as the log2 fold change, is plotted on the x-axis, and the statistical significance, proportional to the negative log10-adjusted p value, is plotted on the y-axis. Yellow and cyan highlight proteins that are statistically significantly up- and downregulated, respectively.*
CS: current smokers, FS: former smokers, NS: never smokers, and eCig: electronic-vapor product users.

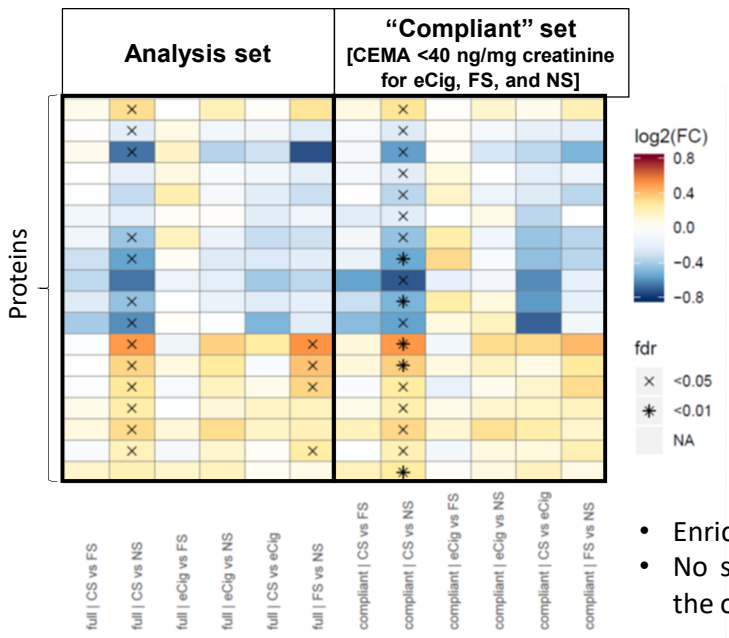
Analysis set
"Compliant" set
[CEMA <40 ng/mg creatinine for eCig, FS, and NS]
Proteins
log2(FC)
0.8
0.4
0.0
−0.4
−0.8
fdr
<0.05
<0.01
NA
full | CS vs FS
full | CS vs NS
full | eCig vs FS
full | eCig vs NS
full | CS vs eCig
full | FS vs NS
compliant | CS vs FS
compliant | CS vs NS
compliant | eCig vs FS
compliant | eCig vs NS
compliant | CS vs eCig
compliant | FS vs NS

Figure 5. Heatmap of the top differentially abundant proteins based on comparisons where a protein is differentially abundant. *For each protein, the heatmap displays the log2 fold change. Differentially abundant proteins are marked with * to indicate a 0.01 FDR (false discovery rate) or x to indicate a 0.05 FDR. CS: current smokers, FS: former smokers, NS: never smokers, and eCig: electronic-vapor product users.*

- Enrichment in immune related proteins for the CS versus NS
- No significant differentially expressed protein detected for the other comparisons

Targeted analysis (PQ500) versus profiling comparison

Figure 6. Venn diagram showing the number of protein groups identified by using either PQ500 (blue) or profiling DIA (red).

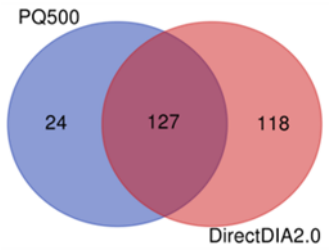
PQ500
24
127
118
DirectDIA2.0

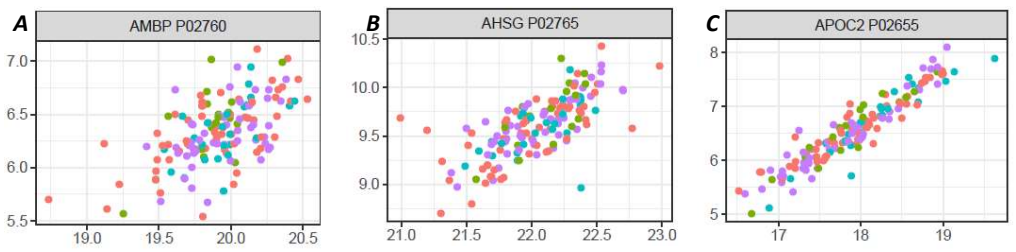
A
AMBP P02760
B
AHSG P02765
C
APOC2 P02655

Figure 7. Example of correlation between the PQ500 and DIA results.
Examples for poor (A), medium (B), and high correlation (C) between the DIA and PQ500 results. No clear trend identified in the dataset.

Predictability model

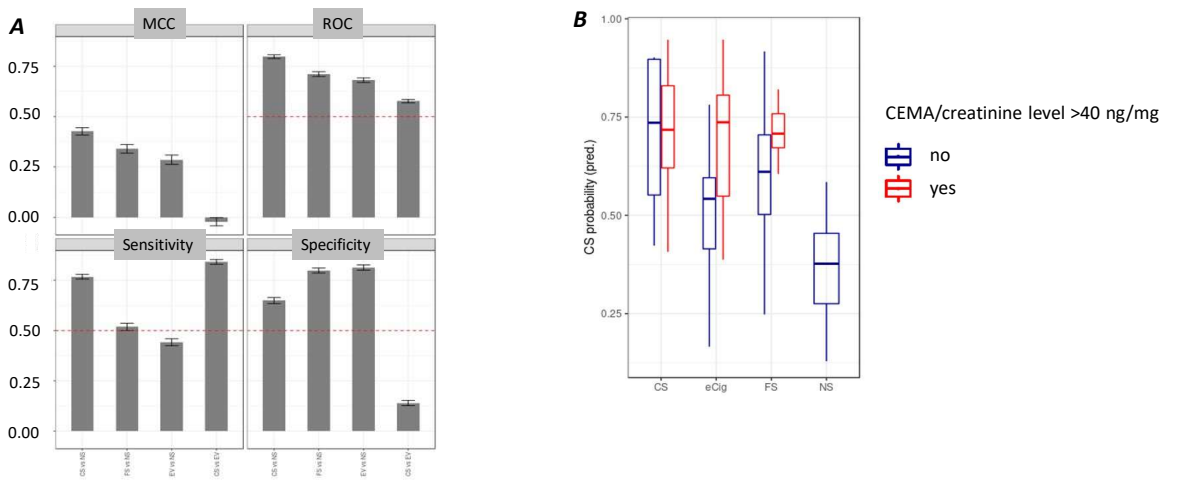
A
MCC
ROC
Sensitivity
Specificity
B
CS probability (pred.)
CEMA/creatinine level >40 ng/mg
no
yes
CS
eCig
FS
NS

Figure 8. A. Predictability in elastic net model based on the analysis set. B Predicted CS probabilities across groups.
CS: current smokers, FS: former smokers, NS: never smokers and eCig: electronic-vapor product users.

Conclusions

- Human plasma study on 204 plasma samples successfully performed:
 - A total of 235 protein groups identified with DIA
 - A total of 151 protein groups identified with PQ500
 - 129 protein groups consistently identified across all samples with DIA
- Subject compliance was assessed by means of CEMA levels in urine. While the CS and NS groups demonstrated the expected separation, CEMA analysis suggested persistent exposure to cigarette smoke in a substantial fraction of the eCig (potentially dual use) and FS groups.
- A new prediction model for subject compliance has been developed.
- Multiomics approaches are planned (metabolomics and transcriptomics) to further complement these results.

PMI SCIENCE
PHILIP MORRIS INTERNATIONAL

HUPO conference

19 Oct – 22 Oct 2020

Competing Financial Interest

All authors are employees of Philip Morris International (PMI) or worked for PMI under contractual agreements. PMI is the sole source of funding and sponsor of this project.