Characterization of aerosol exposure, metabolism and pharmacokinetics for toxicological assessment using in vitro organotypical airway models

A. Knorr, Q. Dutertre, M. Rhouma, E. Martin, M. Almstetter, S. Majeed, C. Mathis, S. Frentzel, M. Bentley, J. Hoeng, P. Vanscheeuwijck, M. C. Peitsch

Philip Morris International R&D, Philip Morris Products S.A., Quai Jeanrenaud 5, 2000 Neuchâtel, Switzerland (part of Philip Morris International group of companies)

Introduction and Objectives

Direct exposure to inhaled mainstream cigarette smoke is known to cause smoking-related damage in the human lung. Aerosol exposure of human three dimensional (3D) organotypical airway epithelial tissue cultures, growing at an air-liquid interface, is a well-established in vitro model enabling a Systems Biology-based Reduced Risk Product (RRP) assessment. Migration kinetics and metabolism of deposited aerosol compounds are important parameters in understanding their bioavailability. To better understand the role of xenobiotic metabolism after cigarette smoking, human subcellular liver and lung fraction models (microsomes, S9) have been established to assess metabolite profiles and reactive metabolites relevant for toxicological assessment. The integration of exposure characterization results with a systems toxicology approach to measure the biological impact of exposure is informative to the product assessment strategy and has the potential to highlight modes of action associated with RRP exposure.

Methods

Aerosol Generation and Exposure of Organotypical Tissue Cultures at the Air-Liquid Interface (ALI)

Smoke Generation | Tissue Exposure System | Cell culture inserts & exposure at ALI | Aerosol/Smoke exposed organotypical tissue cultures

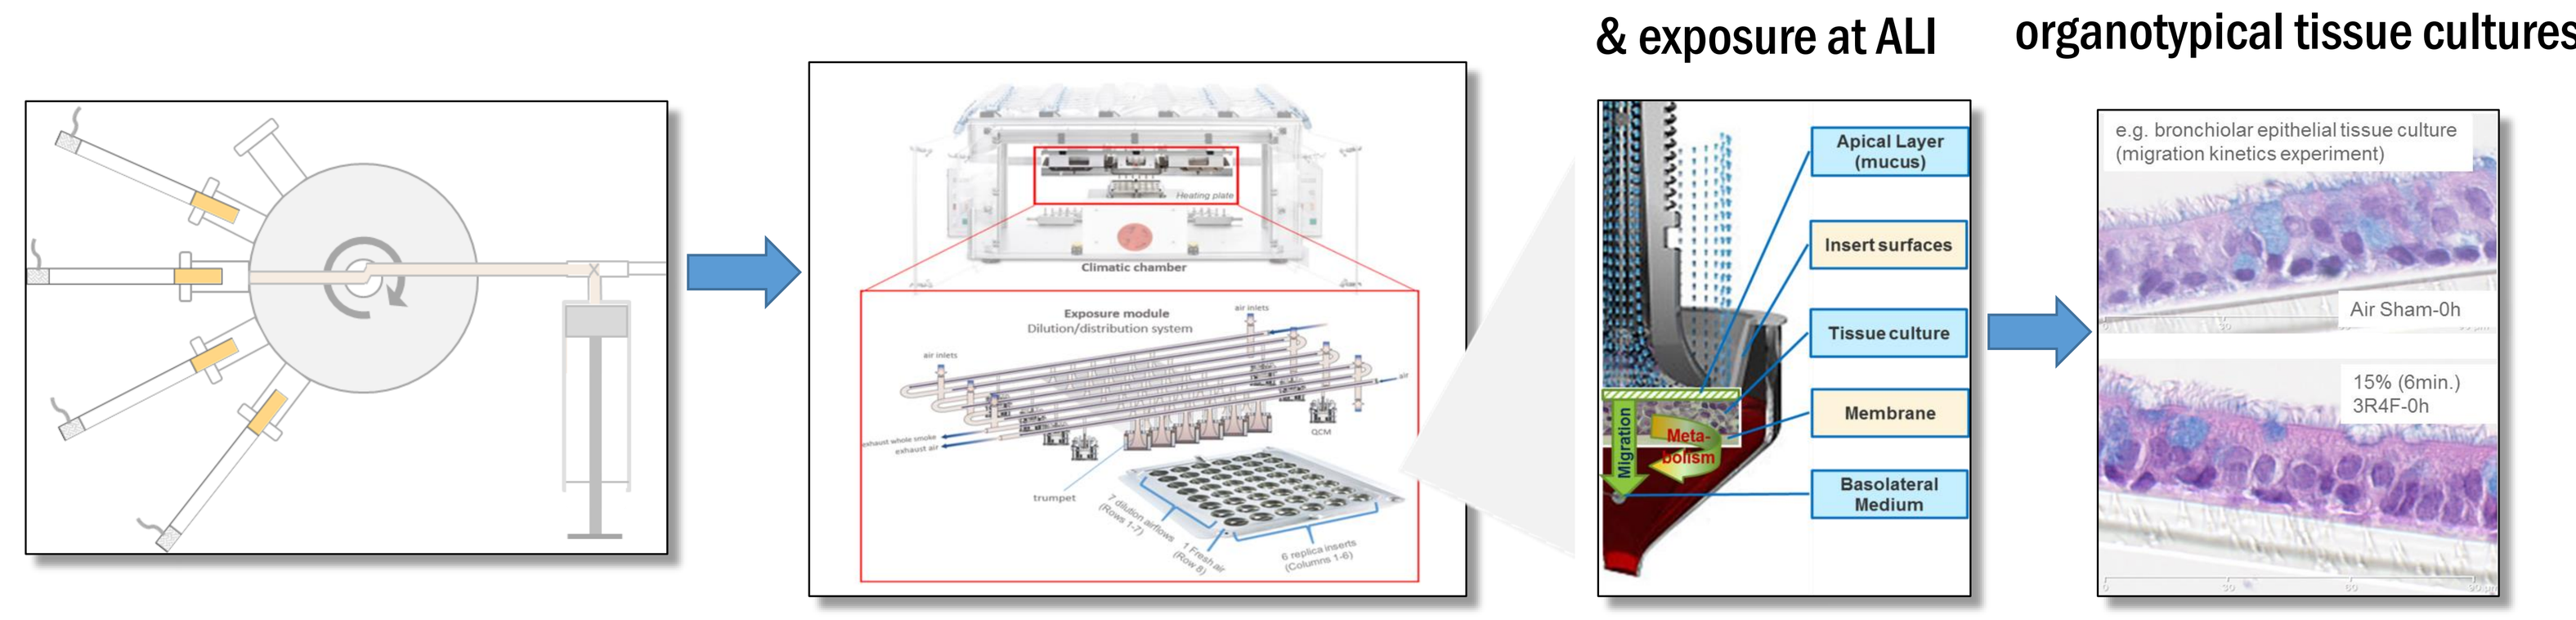
Climatic chamber
Exposure module
Dilution/distribution system
Apical Layer (mucus)
Insert surfaces
Tissue culture
Membrane
Basolateral Medium
Migration
Meta-bolism
e.g. bronchiolar epithelial tissue culture (migration kinetics experiment)
Air Sham-0h
15% (6min.) 3R4F-0h

Smoking machine: SM2000, Philip Morris International
Puff profile: Health Canada regimen (2 puffs/min. of 55 mL and 2 sec. of aspiration and 8 sec. exhaust)

Exposure system: VITROCELL® 24/48 simultaneous exposure of 48 cell culture inserts with up to 7 different smoke dilutions with humidified air and exposing 6 inserts per dilution.

Organotypical Tissue Cultures:
- Bronchiolar, Nasal, Buccal (human)

Surrogate Matrix:
- Phosphate Buffered Saline (PBS)

Figure 1: Aerosol Generation and Exposure of Organotypical Tissue Cultures at the Air-Liquid Interface.

Analytical Methods

Nicotine in Aerosol:
GC-FID after trapping of the aerosol by Extrelut 3NT columns, connected to VITROCELL®

Carbonyls (8) in PBS:
LC-MS/MS after trapping of the aerosol in phosphate buffered saline (PBS) in the wells of the Cultivation base module, derivatization with 2,4-dinitrophenylhydrazine (DNPH)

Aerosol Characterization GC×GC-TOFMS:
2-Dimensional gas chromatography:
- Agilent 7890A + LN2 Modulator + secondary oven
- Injection: Cool-on-column, 0.1µL
- Column 1: 30m DB-5ms
- Column 2: 2.2m DB-17ht

	primary oven	secondary oven
initial	30°C (2min)	35°C (2min)
rate	5°C/min	5.2°C/min
final	320°C (15min)	340°C (14.5min)

Mass Spectrometry (TOFMS):
- LECO Pegasus 4D
- Ionization: EI, 70eV
- Scan range: 35-700 Da
- Data acquisition rate: 200 spectra/s

Characterization of Exposure, Metabolism and Kinetics LC-HRAM-MS:

Traditional method:
- LC-pump: Thermo Accela 1250
- Column: Hypersil GOLD™ aQ (150x2.1 mm, 1.9µ)

Time [min]	A [%]	B [%]
	0.1% formic acid (pH2.7)	0.1% formic acid in acetonitril
0	100	0
3.0	100	0
18.0	10	90
24.0	0	100

- Flow: 400µL/min, 40°C, Inj.: 5µL

New multi-column method:
- Columns:
 1) Biobasic SAX (50x2.1 mm, 5µ)
 2) Kinetex PFP (150x2.1mm, 2.6µ)

Time [min]	A [%]	B [%]
	0.1% formic acid / 10mM NH4formiate (pH3.4)	0.1% formic acid / 1mM NH4formiate in MeOH
0	98	2
4.0	90	10
8.0	60	40
10.0	0	100

- Flow: 250µL/min, 40°C, Inj.: 1.5µL

Mass Spectrometry (HRAM-MS): Thermo QExactiveTM, HESI(+/-)
- Full scan: 80 – 800 Da, res.: 70000
- Full scan: 145 – 650 Da, res.: 70000

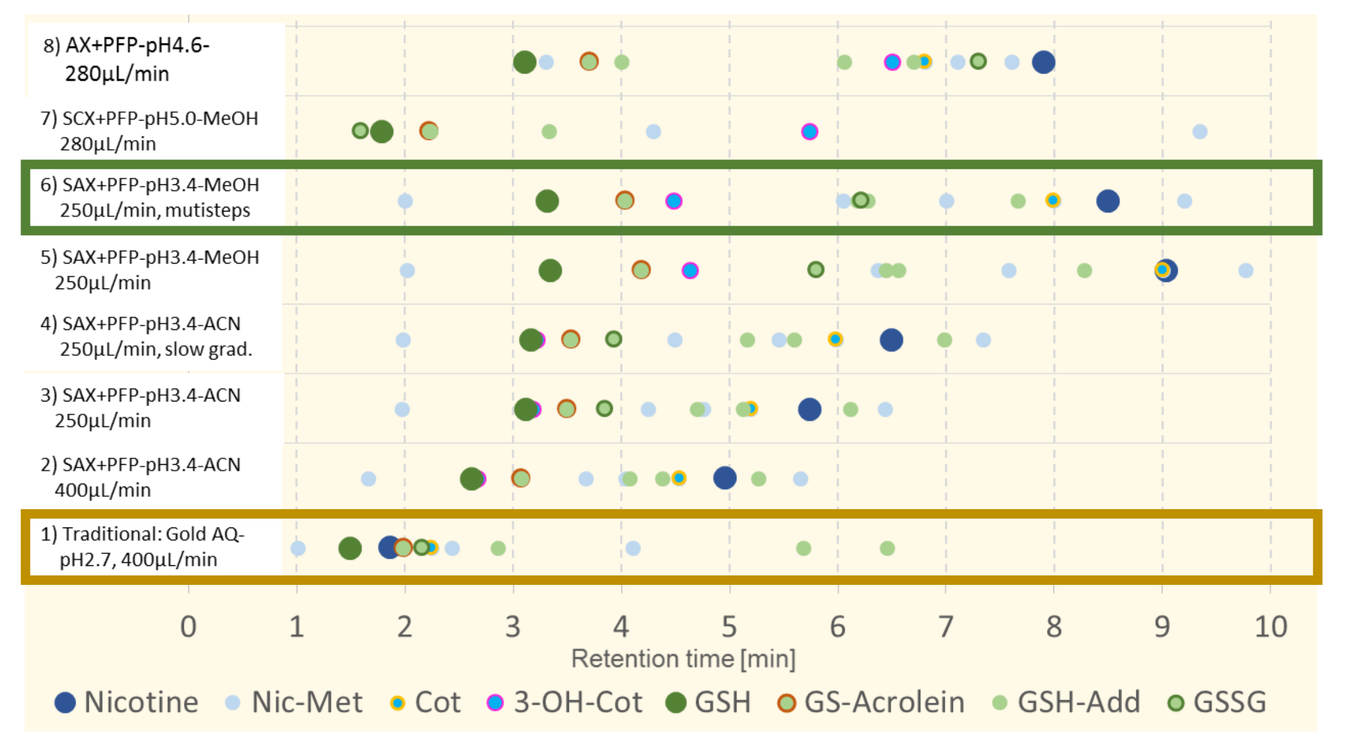
8) AX+PFP-pH4.6-280µL/min
7) SCX+PFP-pH5.0-MeOH 280µL/min
6) SAX+PFP-pH3.4-MeOH 250µL/min, mutisteps
5) SAX+PFP-pH3.4-MeOH 250µL/min
4) SAX+PFP-pH3.4-ACN 250µL/min, slow grad.
3) SAX+PFP-pH3.4-ACN 250µL/min
2) SAX+PFP-pH3.4-ACN 400µL/min
1) Traditional: Gold AQ-pH2.7, 400µL/min
Retention time [min]
Nicotine
Nic-Met
Cot
3-OH-Cot
GSH
GS-Acrolein
GSH-Add
GSSG

Optimized multi-column separation:
- High abundant compounds (e.g. nicotine, GSH, GS-acrolein) well separated from low abundant metabolites (e.g. 3-hydroxycotinine)
- Polar compounds well separated from chromatographic dead-volume

Traditional reversed phase method:
- Poor retention for polar compounds
- Co-elution for most compounds of interest

Figure 2: Chromatographic approaches for separation of nicotine, red./ox. glutathione (GSH/GSSG), GSH adducts of acrolein, crotonaldehyde and benzoquinone and nicotine metabolites (nicotine-N-oxide, cotinine-N-oxide, cotinine, 3-hydroxycotinine, nicotine-N-glucuronide, cotinine-N-glucuronide)

Results

Vitrocell Characterization

Aerosol deposition for the reference cigarette 3R4F and for THS2.2*

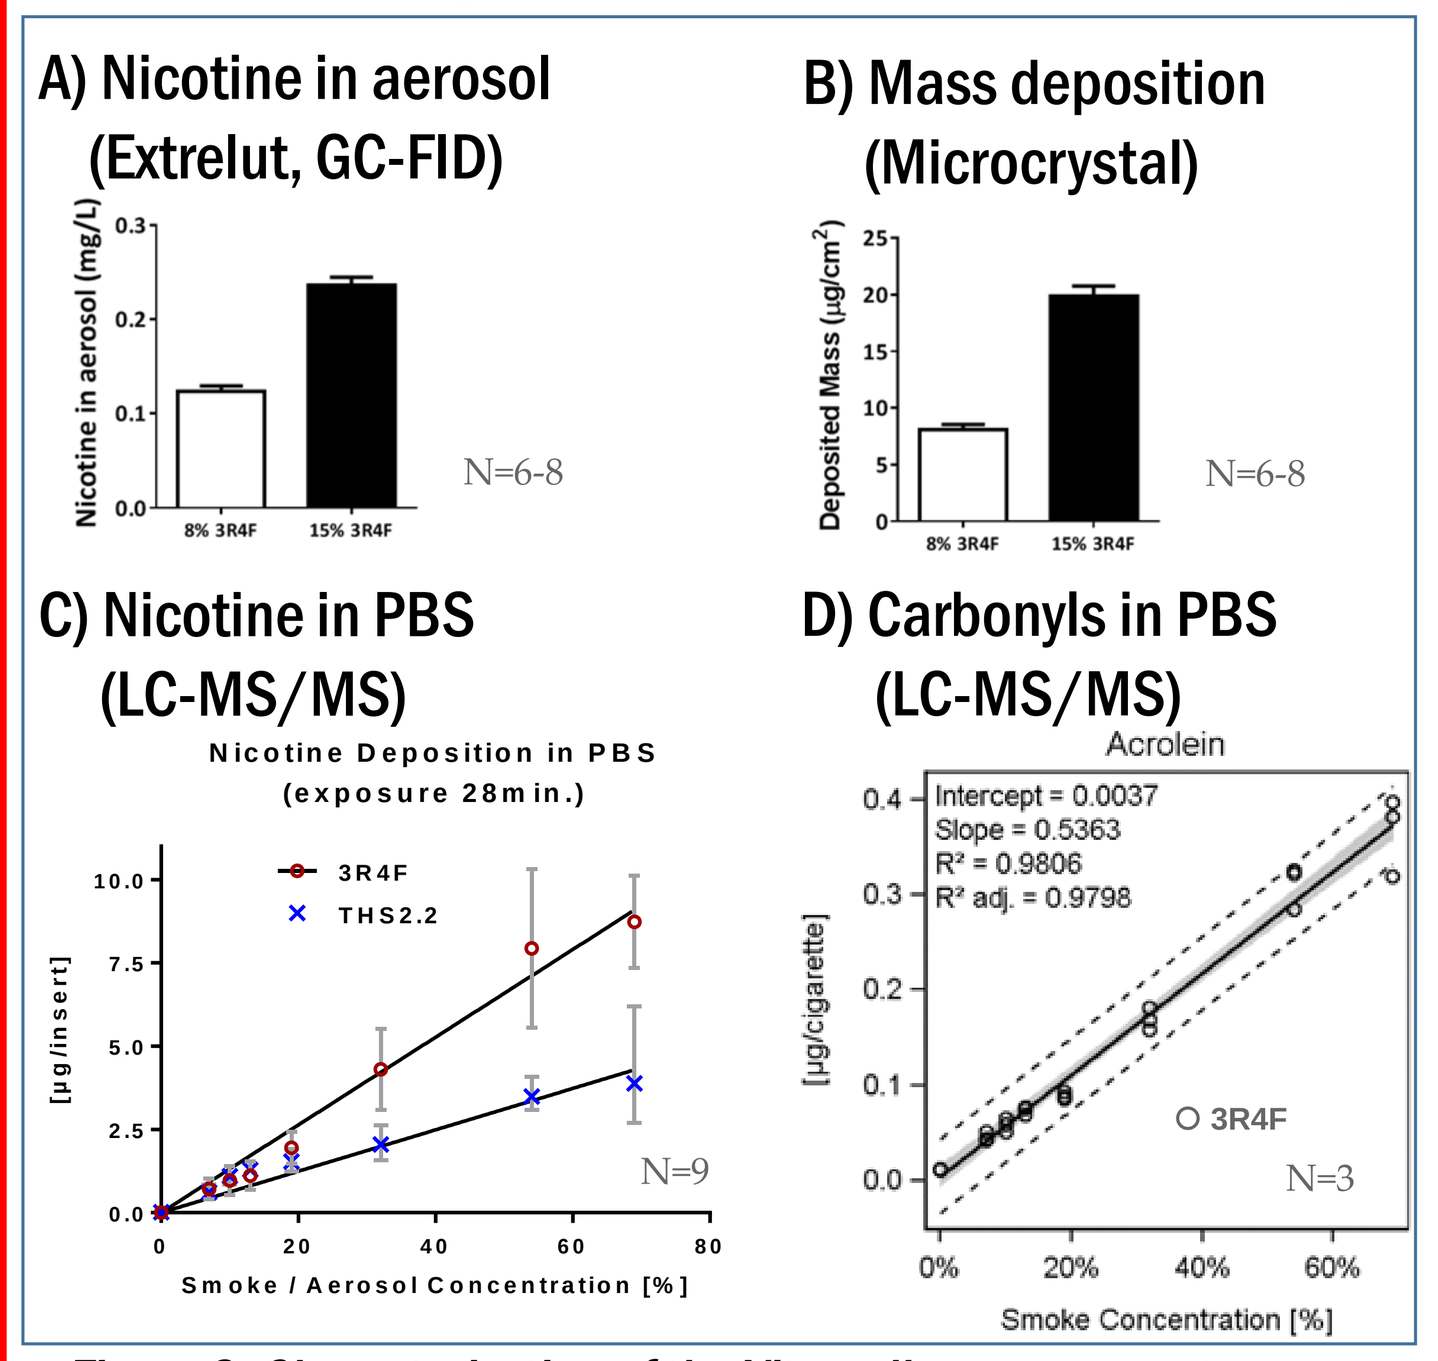
A) Nicotine in aerosol (Extrelut, GC-FID)
Nicotine in aerosol (mg/L)
8% 3R4F
15% 3R4F
N=6-8
B) Mass deposition (Microcrystal)
Deposited Mass (µg/cm²)
N=6-8
C) Nicotine in PBS (LC-MS/MS)
Nicotine Deposition in PBS (exposure 28min.)
3R4F
THS2.2
[µg/insert]
Smoke / Aerosol Concentration [%]
N=9
D) Carbonyls in PBS (LC-MS/MS)
Acrolein
Intercept = 0.0037
Slope = 0.5363
R² = 0.9806
R² adj. = 0.9798
[µg/cigarette]
3R4F
N=3
Smoke Concentration [%]

Figure 3: Characterization of the Vitrocell exposure system: A) nicotine in the aerosol, B) total mass deposited on Quartz Crystal Microbalances (QCM's), C) nicotine deposition in PBS for cigarette whole smoke or THS2.2 aerosol for a complete dose range, D) carbonyls, e.g. acrolein deposition in PBS

Non-targeted screening of smoke deposited at a microcrystal insert (GC×GC-TOFMS)

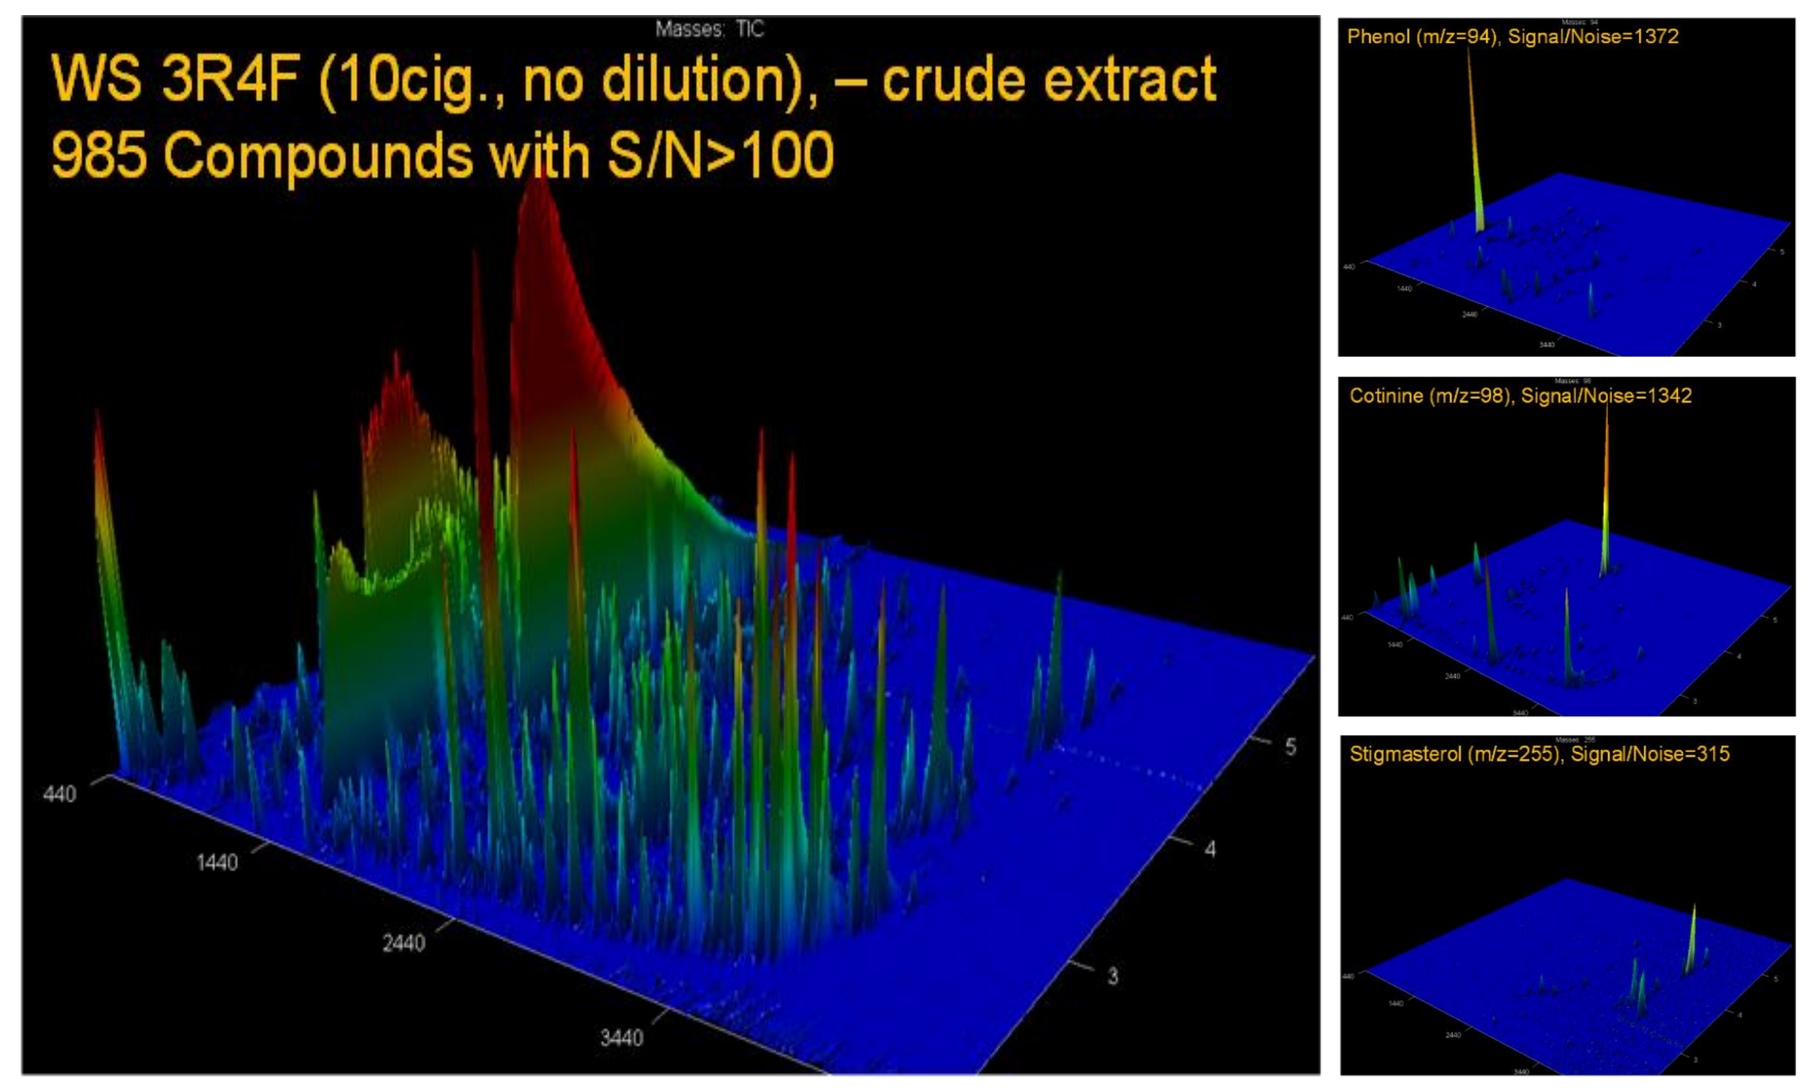
WS 3R4F (10cig., no dilution), – crude extract
985 Compounds with S/N>100
Phenol (m/z=94), Signal/Noise=1372
Cotinine (m/z=98), Signal/Noise=1342
Stigmasterol (m/z=255), Signal/Noise=315

Figure 4: Comprehensive, non-targeted screening of whole smoke (WS) of a reference cigarette 3R4F deposited on a microcrystal insert of a QCM demonstrates the complexity in chemical composition of deposited particles, smoke compounds such as e.g. phenol, cotinine and stigmasterol are shown as an example

**THS2.2 = Tobacco Heating System 2.2*

Exposure Characterization

Exposure of organotypical airway epithelial tissue cultures to aerosol from THS2.2 or smoke from the reference cigarette 3R4F

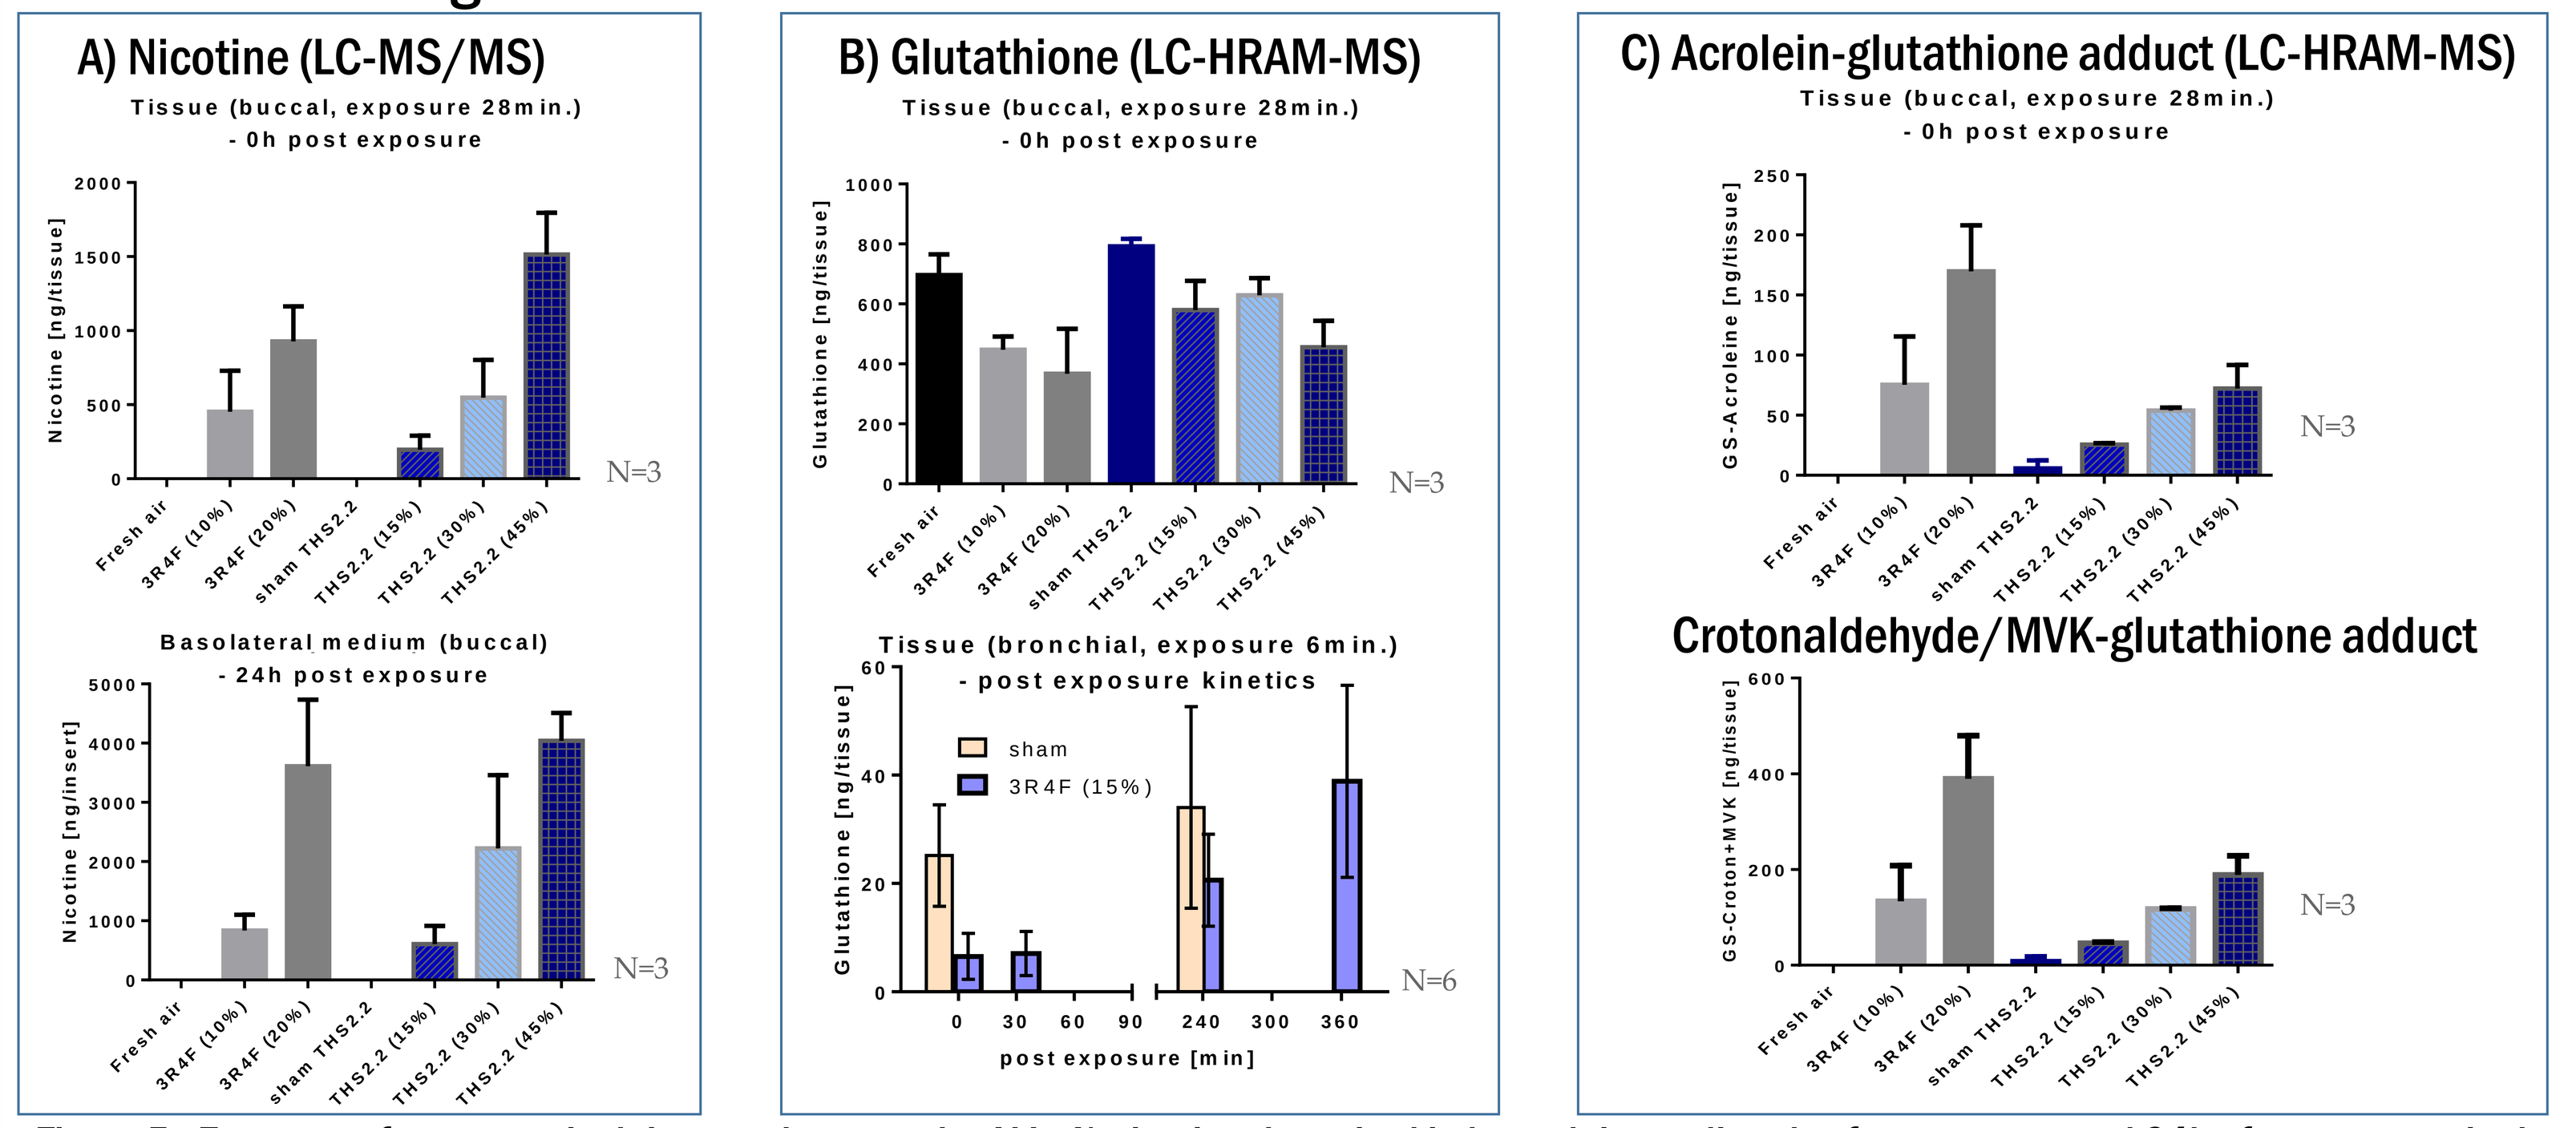
A) Nicotine (LC-MS/MS)
Tissue (buccal, exposure 28min.) - 0h post exposure
Nicotine [ng/tissue]
Basolateral medium (buccal) - 24h post exposure
Nicotine [ng/insert]
Fresh air
3R4F (10%)
3R4F (20%)
sham THS2.2
THS2.2 (15%)
THS2.2 (30%)
THS2.2 (45%)
N=3
B) Glutathione (LC-HRAM-MS)
Glutathione [ng/tissue]
Tissue (bronchial, exposure 6min.) - post exposure kinetics
sham
3R4F (15%)
post exposure [min]
N=6
C) Acrolein-glutathione adduct (LC-HRAM-MS)
GS-Acroleine [ng/tissue]
Crotonaldehyde/MVK-glutathione adduct
GS-Croton+MVK [ng/tissue]

Figure 5: Exposure of organotypical tissue cultures at the ALI, A) nicotine deposited in buccal tissue directly after exposure and 24h after exposure in the basolateral medium, B) glutathione level in exposed tissues for different exposure doses and time points, C) glutathione adducts of electrophilic smoke compounds acrolein and sum of crotonaldehyde and methylvinylketone

Metabolism

Cell-free Model | *In-vitro* Model

Case study: Metabolism of furan-type compounds using human lung and liver microsomes / S9 (cytosolic fraction)

Analytical challenge: multiplicity of initial adducts is formed, which are also subject to subsequent metabolic reduction

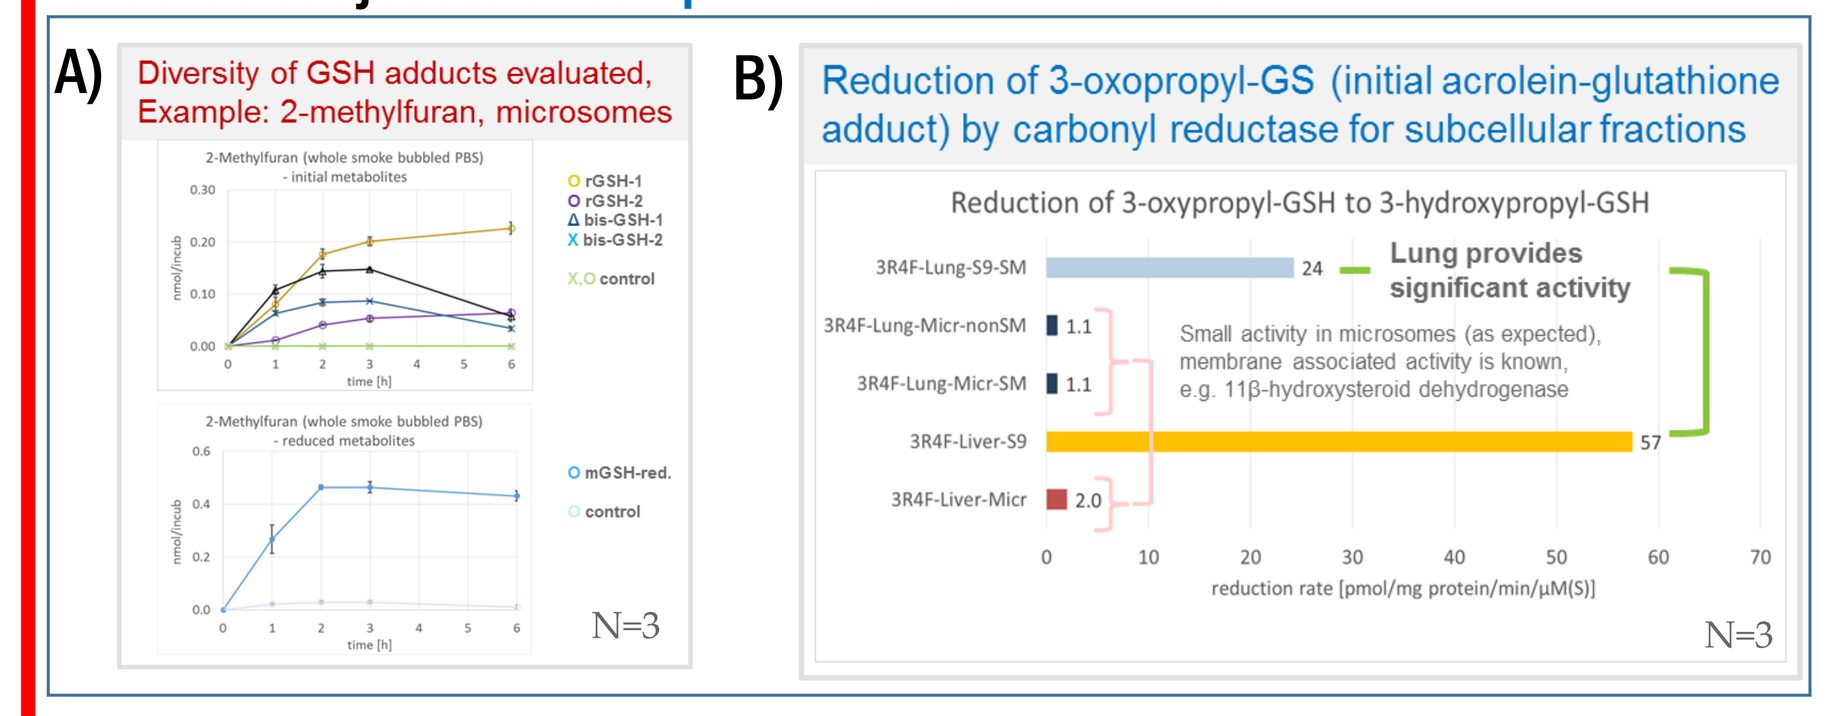
A) Diversity of GSH adducts evaluated, Example: 2-methylfuran, microsomes
N=3
B) Reduction of 3-oxopropyl-GS (initial acrolein-glutathione adduct) by carbonyl reductase for subcellular fractions
Reduction of 3-oxypropyl-GSH to 3-hydroxypropyl-GSH
3R4F-Lung-S9-SM
3R4F-Lung-Micr-nonSM
3R4F-Lung-Micr-SM
3R4F-Liver-S9
3R4F-Liver-Micr
Lung provides significant activity
Small activity in microsomes (as expected), membrane associated activity is known, e.g. 11β-hydroxysteroid dehydrogenase
reduction rate [pmol/mg protein/min/µM(S)]
N=3

Figure 6: A) Bioactivation of furan and simple alkylfurans followed by conjugation to glutathione and subsequent metabolic reduction. B) Rate of reduction of the initial acrolein glutathione adduct for different subcellular fractions

Results of furans case study (summary):
- Human lung microsomes provide CYP2E1 activity
- Human lung microsomes are able to detoxify furan(s) (CYP2E1)
- Human lung S9 generates reactive sulfates of hydroxymethyl-furans (e.g. 5-HMF)

Human organotypical tissue cultures exposed to whole smoke from the reference cigarette 3R4F

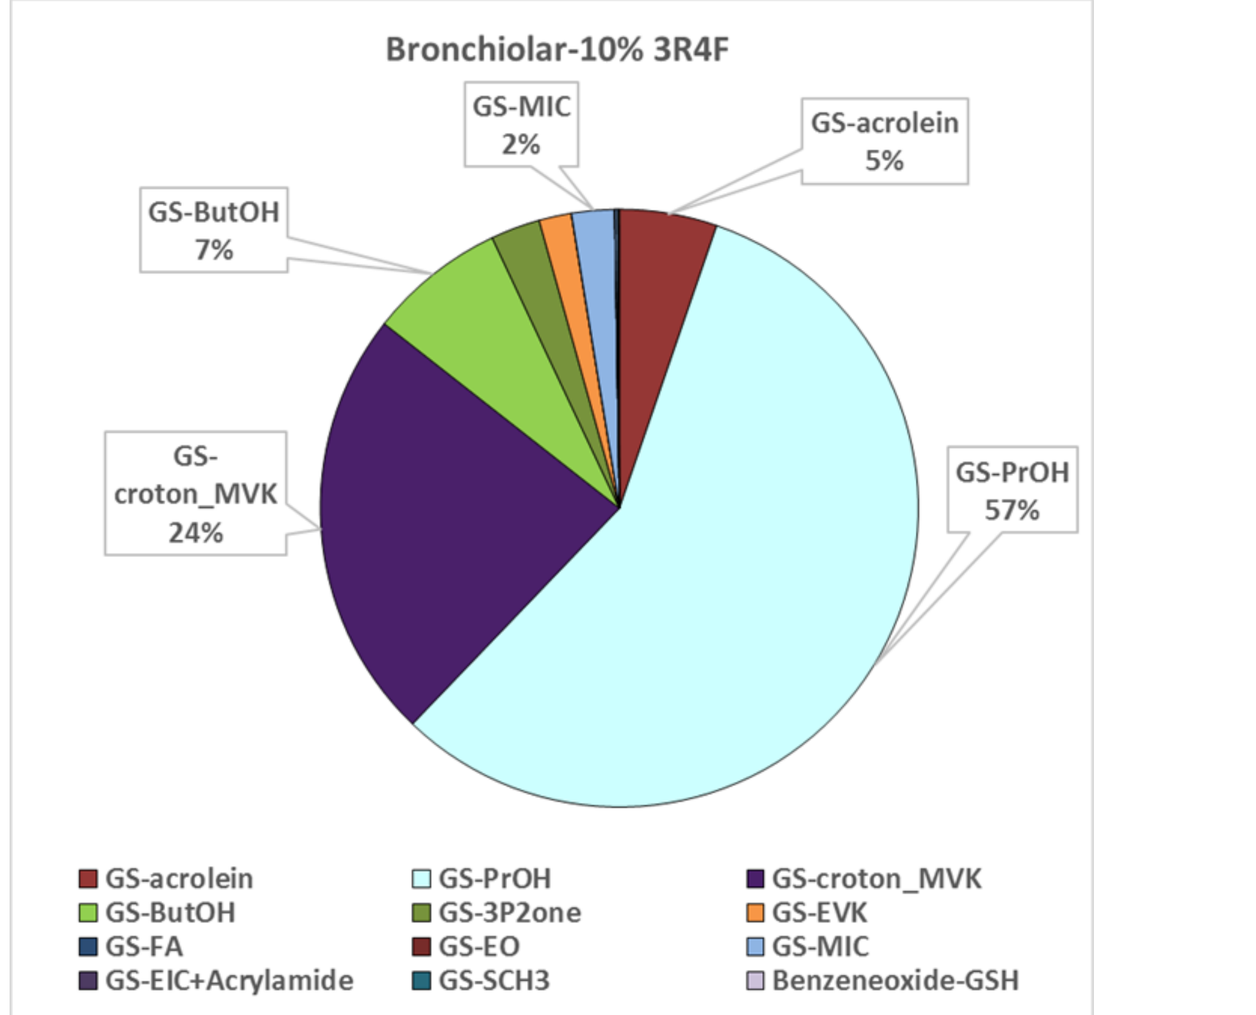
Bronchiolar-10% 3R4F
GS-MIC 2%
GS-acrolein 5%
GS-ButOH 7%
GS-croton_MVK 24%
GS-PrOH 57%
GS-acrolein
GS-ButOH
GS-FA
GS-EIC+Acrylamide
GS-PrOH
GS-3P2one
GS-EO
GS-SCH3
GS-croton_MVK
GS-EVK
GS-MIC
Benzeneoxide-GSH

Figure 7: Acrolein (measured by GS-acrolein as initial adduct, see Figure 6B and GS-PrOH as metabolic reduction product), crotonaldehyde and MVK[1] are responsible for the majority (approx. 92-93%) of the GSH-adducts identified

[1] *MVK: methylvinylketone*

Migration Kinetics

Human organotypical bronchiolar tissue cultures exposed to cigarette smoke

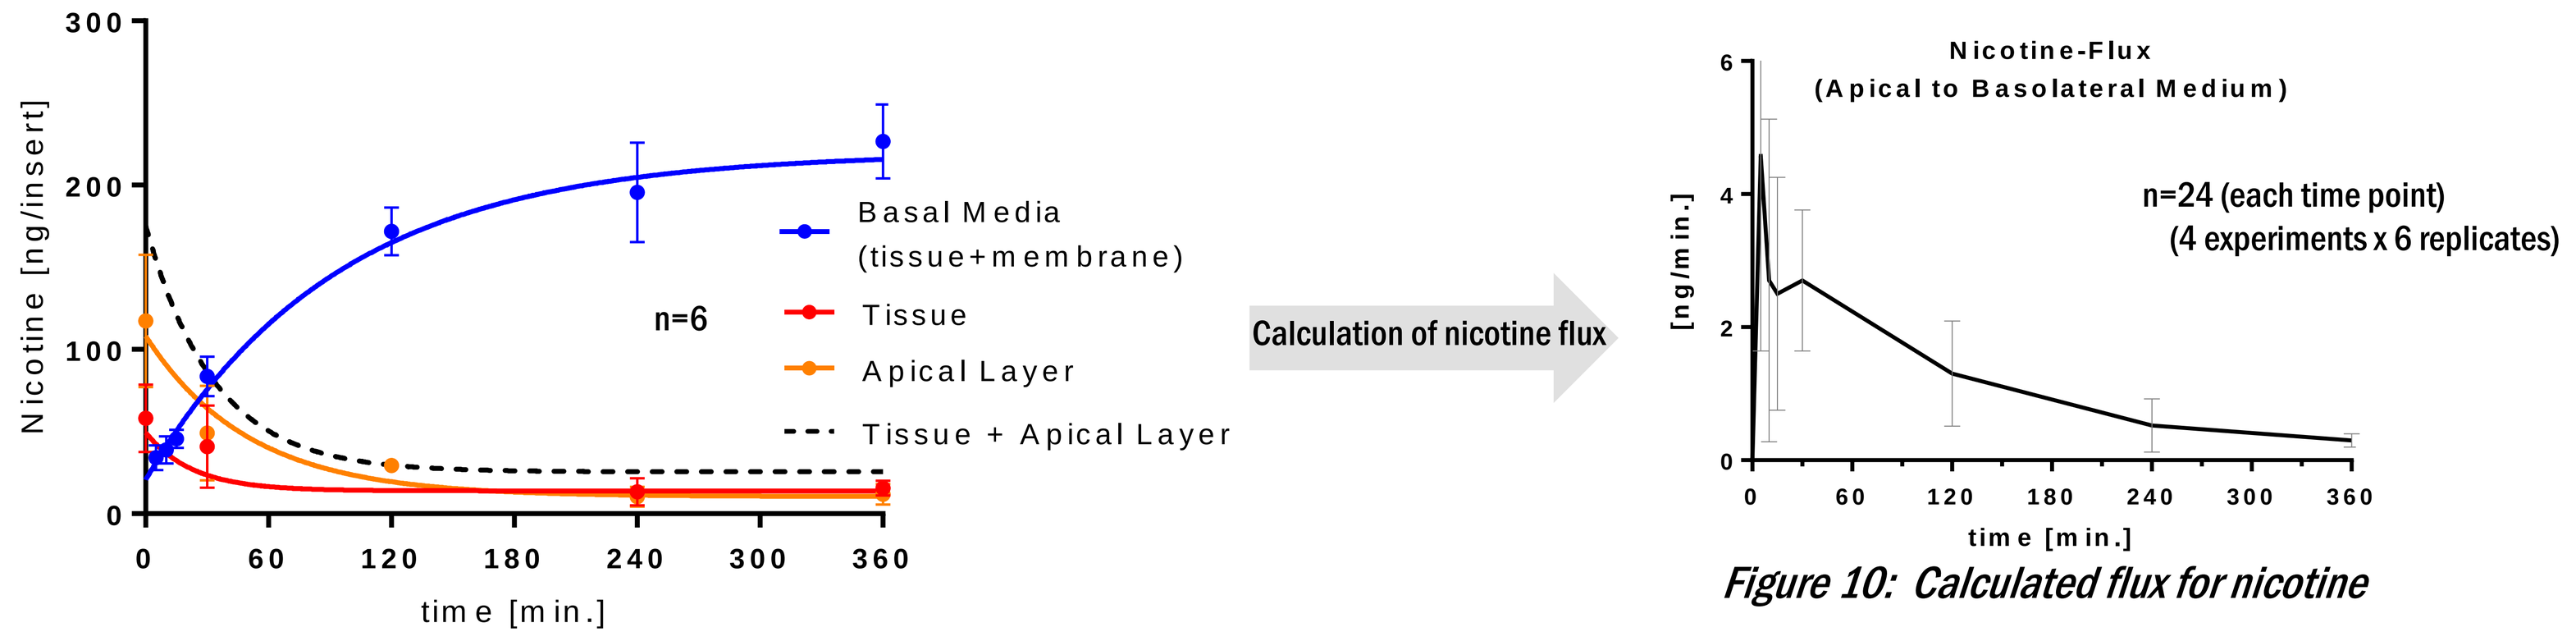
Nicotine [ng/insert]
Basal Media (tissue+membrane)
Tissue
Apical Layer
Tissue + Apical Layer
n=6
time [min.]
Calculation of nicotine flux
Nicotine-Flux (Apical to Basolateral Medium)
[ng/min.]
n=24 (each time point) (4 experiments x 6 replicates)

Figure 8: Migration kinetics for nicotine in bronchiolar tissue, exposed to 15% 3R4F for 6min.

Figure 10: Calculated flux for nicotine

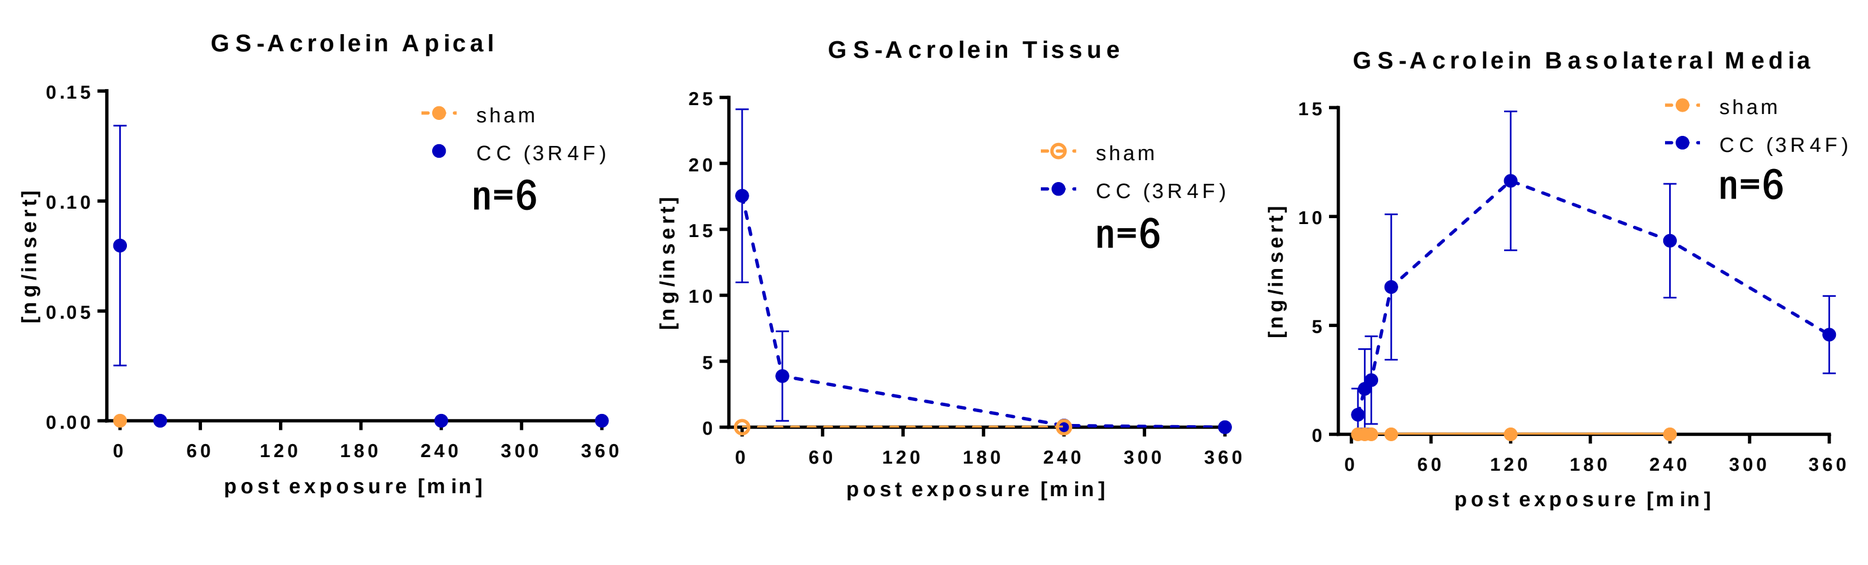
GS-Acrolein Apical
GS-Acrolein Tissue
GS-Acrolein Basolateral Media
sham
CC (3R4F)
n=6
[ng/insert]
post exposure [min]

Figure 9: Kinetics for acrolein glutathione adduct in bronchiolar tissue, exposed to 15% 3R4F for 6min.

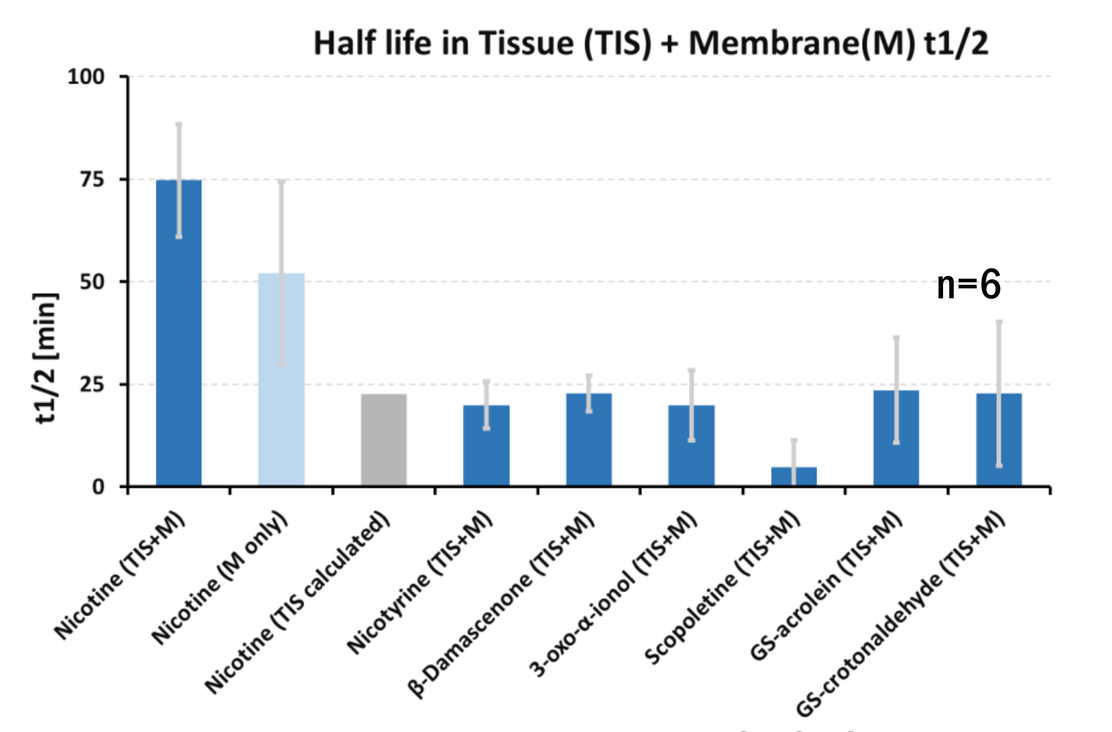
Half life in Tissue (TIS) + Membrane(M) t1/2
t1/2 [min]
n=6
Nicotine (TIS+M)
Nicotine (M only)
Nicotine (TIS calculated)
Nicotyrine (TIS+M)
β-Damascenone (TIS+M)
3-oxo-α-ionol (TIS+M)
Scopoletine (TIS+M)
GS-acrolein (TIS+M)
GS-crotonaldehyde (TIS+M)

Figure 11: Calculated tissue half-life for nicotine and additional compounds

Conclusions

- Subcellular fractions (microsomes, S9) are useful tools to study in vitro lung metabolism:
 - Metabolite profiles and reactive metabolites are relevant for toxicology & kinetics
 - Results from metabolism models using subcellular fractions can be utilized in more complex models, such as organotypical tissue cultures
- Metabolism studies will further substantiate the use of organotypical tissue culture models as a tool for in vitro respiratory toxicology testing
- Analytical methodologies were established to characterize the exposure of organotypical tissue cultures:
 - Nicotine and glutathione adducts of reactive carbonyls can be determined to describe the dose in exposed tissues
 - Glutathione serves as an additional marker for tissue functionality
- Dynamics of the exposed organtypical tissue cultures help to understand the biological system:
 - Migration kinetics (including adsorption, metabolism, excretion kinetics): => bioavailability of aerosol (compounds) by area under the curve (AUC)

References

1) Majeed et al. Characterization of the Vitrocell® 24/48 in vitro aerosol exposure system using mainstream cigarette smoke, Chemistry Central Journal 8(1):62 · December 2014
2) Peterson L. A. Reactive metabolites in the biotransformation of molecules containing a furan ring, Chem. Res. Toxicol. 26:6 (2013).
3) Takakusa H. et al. Markers of Electrophilic Stress Caused by Chemically Reactive Metabolites in Human Hepatocytes, Drug Metabolism and Disposition. Vol. 36, No. 5 (2008).
4) Jose´ V. Castell et al. Metabolism and bioactivation of toxicants in the lung. The in vitro cellular approach, Experimental and Toxicologic Pathology 57 (2005) 189–204
5) University of Kentucky (www.3r4f.com), (http://www2.ca.uky.edu/refcig/3R4F%20Preliminary%20Analysis.pdf)

PMI SCIENCE
PHILIP MORRIS INTERNATIONAL
www.pmiscience.com

64th ASMS Conference on Mass Spectrometry and Allied Topics
San Antonio, TX, USA
June 5 – 9, 2016

Competing Financial Interest
The research described in this poster was funded by Philip Morris International