Systems toxicology analysis of cardiovascular and respiratory endpoints from Apoe$^{-/-}$ mice showed similar effects after switching to a candidate modified risk tobacco product, THS 2.2, or to smoking cessation

Blaine Phillips[a], Emilija Veljkovic[b], Stéphanie Boué[b], Walter K. Schlage[c], Gregory Vuillaume[b], Florian Martin[b], Bjoern Titz[b], Patrice Leroy[b], Ansgar Buettner[d], Ashraf Elamin[b], Alberto Oviedo[a], Maciej Cabanski[b,e], Emmanuel Guedj[b], Thomas Schneider[b], Marja Talikka[b], Nikolai V. Ivanov[b], Patrick Vanscheeuwijck[b], Manuel C. Peitsch[b], Julia Hoeng[b]

a Philip Morris International Research Laboratories Pte Ltd, 50 Science Park Road, The Kendall #02-07, Science Park II, Singapore 117406; b Philip Morris International R&D, Philip Morris Products S.A., Quai Jeanrenaud 5, 2000 Neuchâtel, Switzerland;
c Biology Consultant, Max-Baermann-Str. 21, 51429 Bergisch Gladbach, Germany; d Histovia GmbH, Schöne Aussicht 5, 51491 Overath, Germany; e Current address for Maciej Cabanski: Novartis Pharma AG, Novartis Institutes for Biomedical Research (NIBR), CH-4002, Basel, Switzerland

ABSTRACT

Cigarette smoking is a risk factor for chronic obstructive pulmonary disease (COPD) and cardiovascular disease (CVD). Apoe-deficient mice are prone to developing premature atherosclerosis and emphysema making it an ideal model in which both pathologies can be assessed simultaneously. We evaluated the effects of cigarette smoke (CS) from a conventional cigarette (3R4F) and from THS 2.2, a modified risk tobacco product (MRTP). Apoe$^{-/-}$ mice were exposed for up to 8 months to an aerosol from 3R4F or from (THS 2.2) 3 hours/day, 5 days/week to a target nicotine concentration of 30 µg/l. After 2 months exposure to CS, cessation and switching groups were further exposed for up to 6 months to fresh air, or THS 2.2, respectively. A battery of markers of disease progression were investigated including atherosclerotic plaque formation, pulmonary inflammation (cell infiltration, cytokine levels), pulmonary function and lung emphysema. Exposure to CS induced time-dependent molecular, physiological and inflammatory responses in the lungs of Apoe$^{-/-}$ mice consistent with emphysematous changes. The size of atherosclerotic plaques measured in the aortic arches was higher in CS-exposed animals compared to both sham and pMRTP-exposed animals at all time-points examined. Micro computed tomographic (micro CT) analysis of aortic images after 8 months of exposure showed that larger plaque volumetric measurements in 3R4F-exposed animals than sham-exposed, cessation or MRTP-exposed mice. Significant changes in the lung transcriptome of Apoe$^{-/-}$ mice were observed in response to 3R4F-exposure compared to the gene expression levels observed in the transcriptome of sham-exposed mice. Smoking cessation and switching to THS2.2 resulted in lower activation levels compared to 3R4F. Both, smoking cessation and switching to the MRTP aerosol halted the rate of disease development as assessed by inflammatory, histopathological and molecular endpoints, typically within 1 to 3 months post exposure. Our work demonstrates the power of using the Apoe$^{-/-}$ mouse model to study comorbidities associated with cigarette smoking and investigate the mechanisms underlying the benefits of smoking cessation or switching to a MRTP.

CONCLUSIONS

- The exposure to 3R4F cigarette smoke resulted in significant levels of pulmonary inflammation, decline in pulmonary function, and histopathological changes. These phenotypic changes were coherent with the molecular data.
- Chronic exposure to an aerosol from the THS2.2 resulted in very little difference in all measured parameters related to COPD and CVD when compared to the filtered air-exposed animals.
- The biological response to switching to a THS2.2 aerosol or filtered air following 2 months of 3R4F cigarette smoke exposure were very similar between the two conditions across the spectrum of endpoints assessed, and showed a generally positive effect.
- Differential 'omics' profiles associated with 3R4F exposure returned to nearly filtered air-like level following either switching to a THS2.2 aerosol or filtered air.
- Histopathological assessment also showed a marked effect of switching, in which a partial or complete (depending on the inflammatory cell type) reversal of pulmonary inflammation was observed.
- These data collectively indicate a halting or regression of the disease genesis following switching from conventional cigarette to THS2.2 aerosol in Apoe$^{-/-}$.

TEST ITEMS

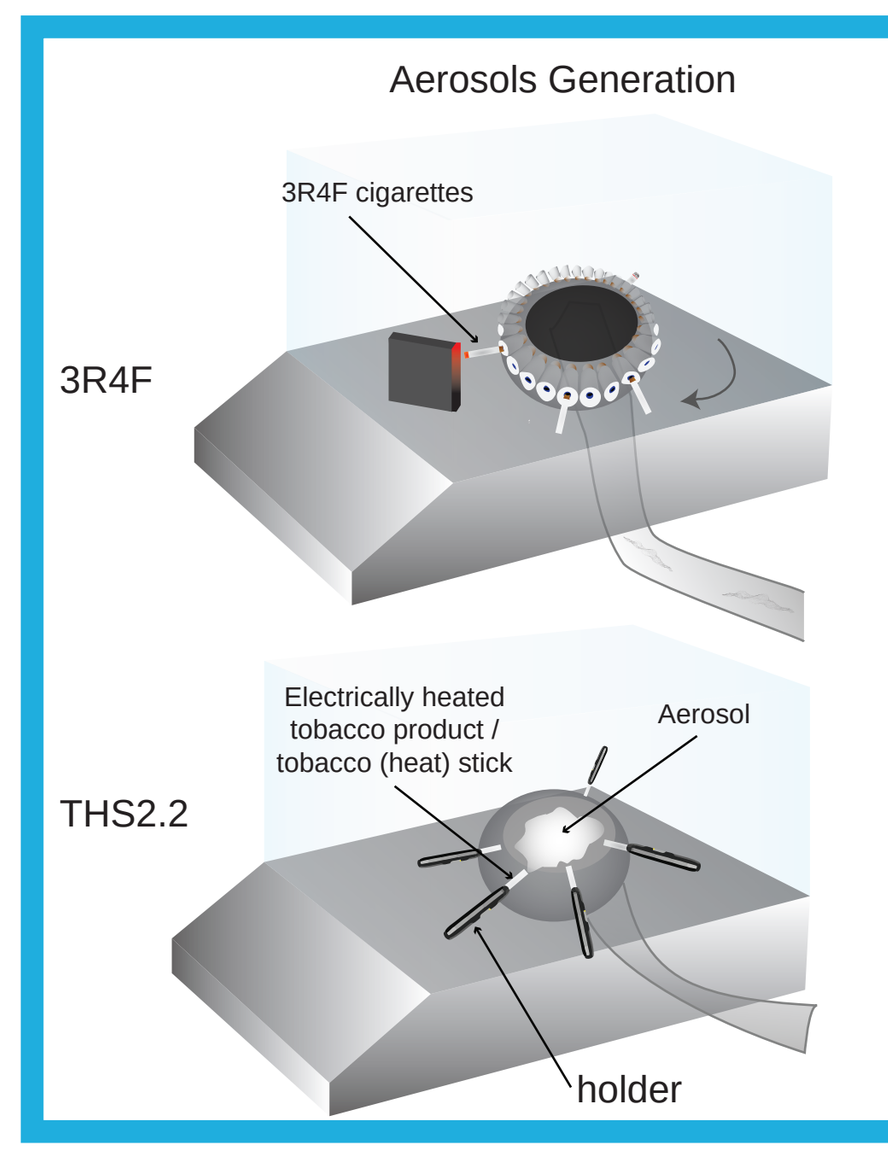
Aerosols Generation
3R4F cigarettes
3R4F
Electrically heated tobacco product / tobacco (heat) stick
Aerosol
THS2.2
holder

3R4F reference cigarettes were purchased from the University of Kentucky (http://www.ca.uky.edu/refcig). The candidate MRTP, THS2.2, consisted of a stick containing a tobacco plug inserted into a holder that heats the tobacco in a controlled way to ensure combustion temperatures are not reached. The controlled heating of the tobacco generates an aerosol containing mainly water, glycerin, nicotine, and tobacco flavors.

Yields per cigarette of selected 3R4F mainstream smoke and THS aerosol constituents (HPHCs), normalized for nicotine levels.

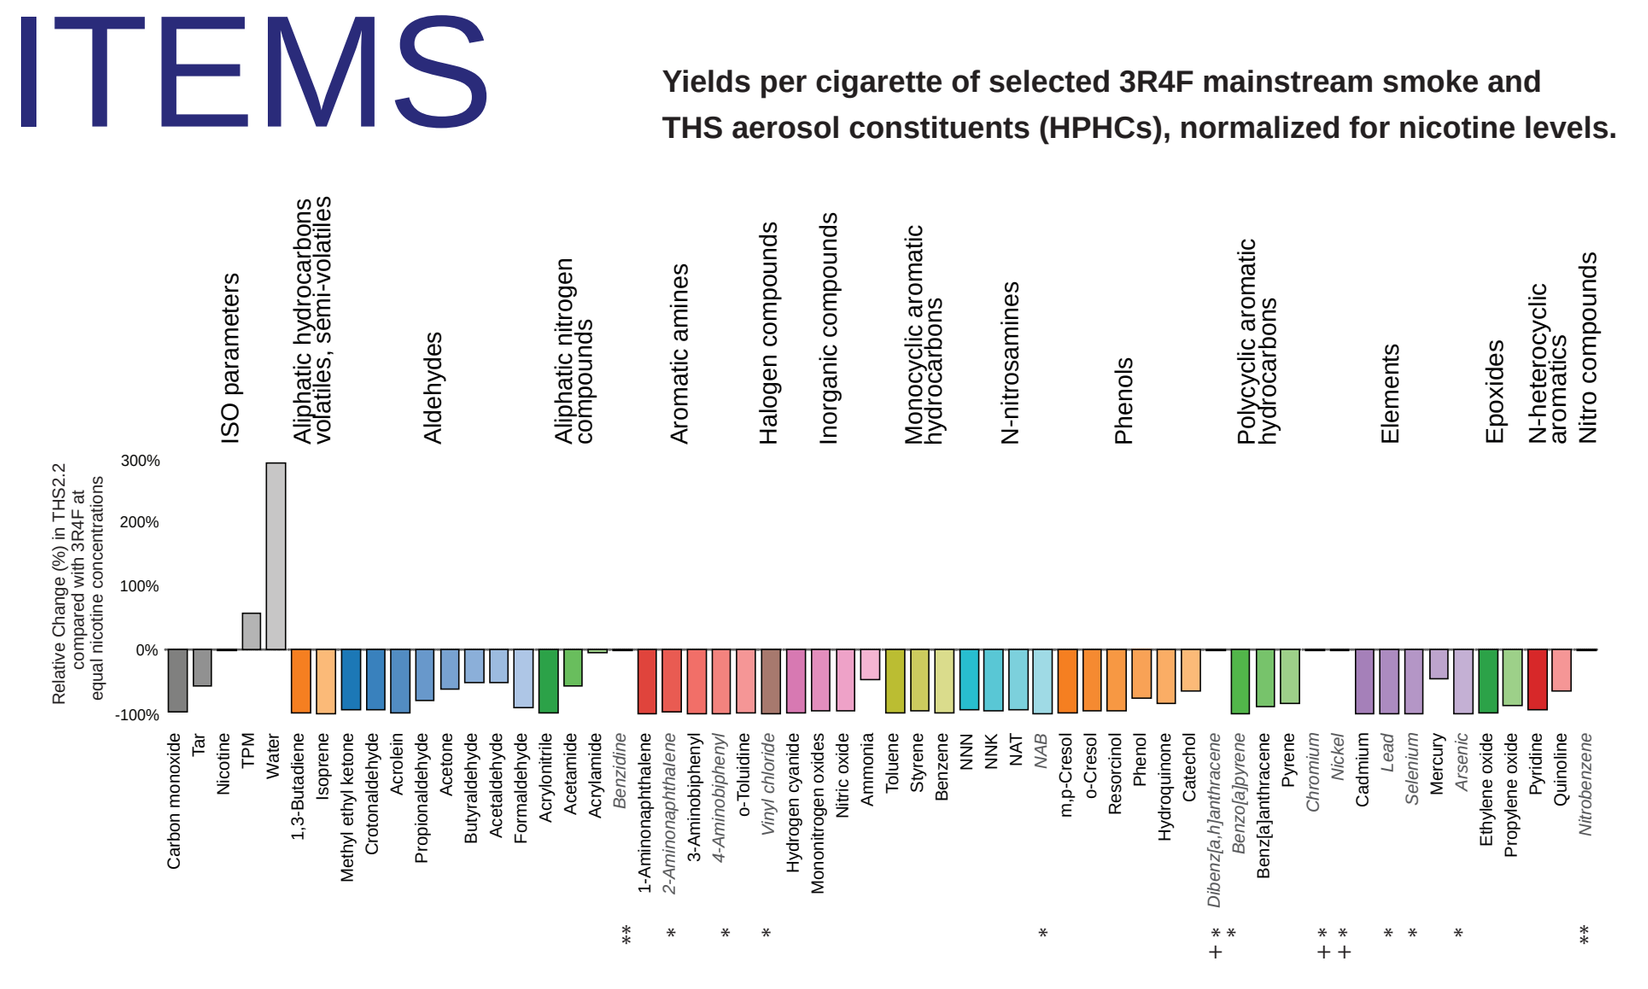
ISO parameters
Aliphatic hydrocarbons volatiles, semi-volatiles
Aldehydes
Aliphatic nitrogen compounds
Aromatic amines
Halogen compounds
Inorganic compounds
Monocyclic aromatic hydrocarbons
N-nitrosamines
Phenols
Polycyclic aromatic hydrocarbons
Elements
Epoxides
N-heterocyclic aromatics
Nitro compounds
Relative Change (%) in THS2.2 compared with 3R4F at equal nicotine concentrations

* <LOQ in THS2.2, replaced by LOQ + <LOQ in 3R4F, replaced by LOQ ** not determined in THS2.2 nor 3R4F

REFERENCES

- Phillips, B., et al. (2015) An 8-Month Systems Toxicology Inhalation/Cessation Study in Apoe-/- Mice to Investigate Cardiovascular and Respiratory Exposure Effects of a Candidate Modified Risk Tobacco Product, THS 2.2, Compared with Conventional Cigarettes., Toxicological Sciences, In press.
- Boue, S., et al. (2013) Cigarette smoke induces molecular responses in respiratory tissues of ApoE-/- mice that are progressively deactivated upon cessation Toxicology, 314, 112 - 124.
- Boue, S., et al. (2015) Causal biological network database: a comprehensive platform of causal biological network models focused on the pulmonary and vascular systems, Database : the journal of biological databases and curation, 2015, bav030.
- Boue, S., et al. (2012) Modulation of atherogenic lipidome by cigarette smoke in apolipoprotein E-deficient mice. Atherosclerosis.
- Food and Drug Administration, F. (2012) Modified Risk Tobacco Product Applications: Draft Guidance for Industry.
- Hoeng, J., et al. (2012) A network-based approach to quantifying the impact of biologically active substances, Drug discovery today, 17, 413-418.
- Kogel, U., et al. (2014) A 28-day rat inhalation study with an integrated molecular toxicology endpoint demonstrates reduced exposure effects for a prototypic modified risk tobacco product compared with conventional cigarettes, Food and chemical toxicology : an international journal published for the British Industrial Biological Research Association, 68, 204-217.
- Lietz, M., et al. (2013) Cigarette-smoke-induced atherogenic lipid profiles in plasma and vascular tissue of apolipoprotein E-deficient mice are attenuated by smoking cessation, Atherosclerosis, 229, 86-93.
- Martin, F., et al. (2014) Quantification of biological network perturbations for mechanistic insight and diagnostics using two-layer causal models, BMC bioinformatics, 15, 238.
- Phillips, B., et al. (2015) A 7-month cigarette smoke inhalation study in C57BL/6 mice demonstrates reduced lung inflammation and emphysema following smoking cessation or aerosol exposure from a prototypic modified risk tobacco product, Food and chemical toxicology : an international journal published for the British Industrial Biological Research Association, 80 328-345.
- Schorp, M.K., Tricker, A.R. and Dempsey, R. (2012) Reduced exposure evaluation of an Electrically Heated Cigarette Smoking System. Part 1: Non-clinical and clinical insights, Regulatory toxicology and pharmacology : RTP, 64, S1-10.
- Sturla, S.J., et al. (2014) Systems toxicology: from basic research to risk assessment, Chemical research in toxicology, 27, 314-329.

LEGEND

- Sham
- 3R4F
- THS2.2
- Cessation
- Switch

*: different from sham (p<0.05)
#: different from 3R4F (p<0.05)
&: different from Cessation (p<0.05)

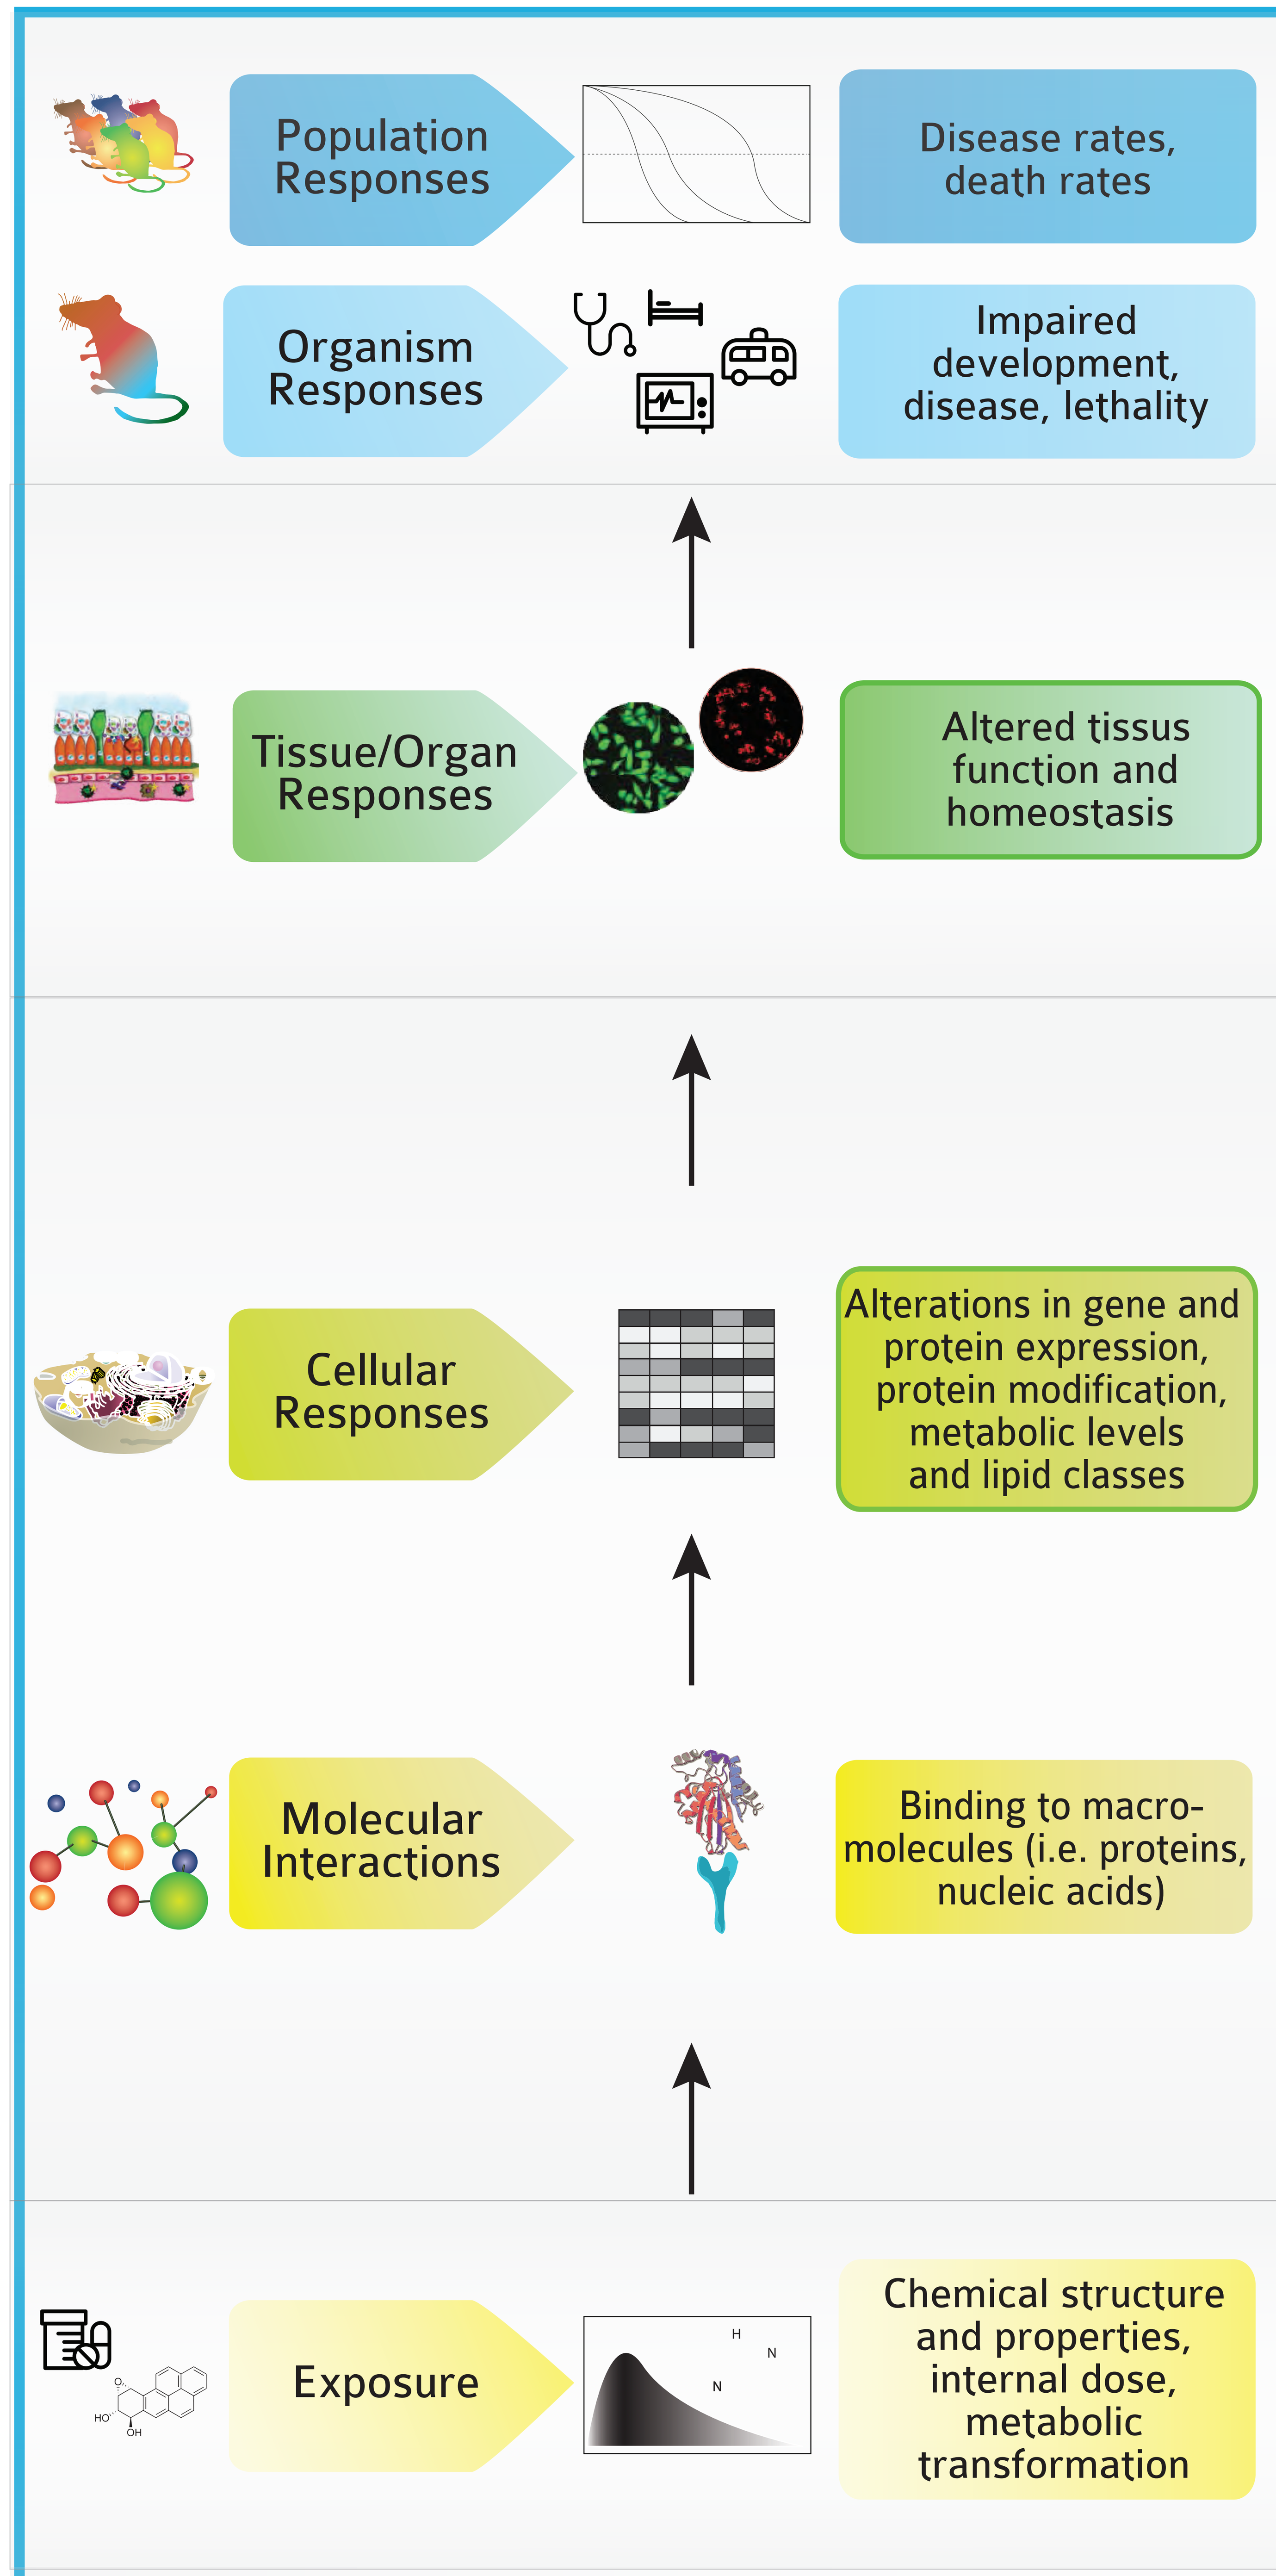
Population Responses
Disease rates, death rates
Organism Responses
Impaired development, disease, lethality
Tissue/Organ Responses
Altered tissus function and homeostasis
Cellular Responses
Alterations in gene and protein expression, protein modification, metabolic levels and lipid classes
Molecular Interactions
Binding to macro-molecules (i.e. proteins, nucleic acids)
Exposure
Chemical structure and properties, internal dose, metabolic transformation

Both 3R4F CS and THS2.2 aerosols were well tolerated by the mice. The overall mortality rate was 3.5% and was not exposure-related (dead/moribund mice spread across all groups).

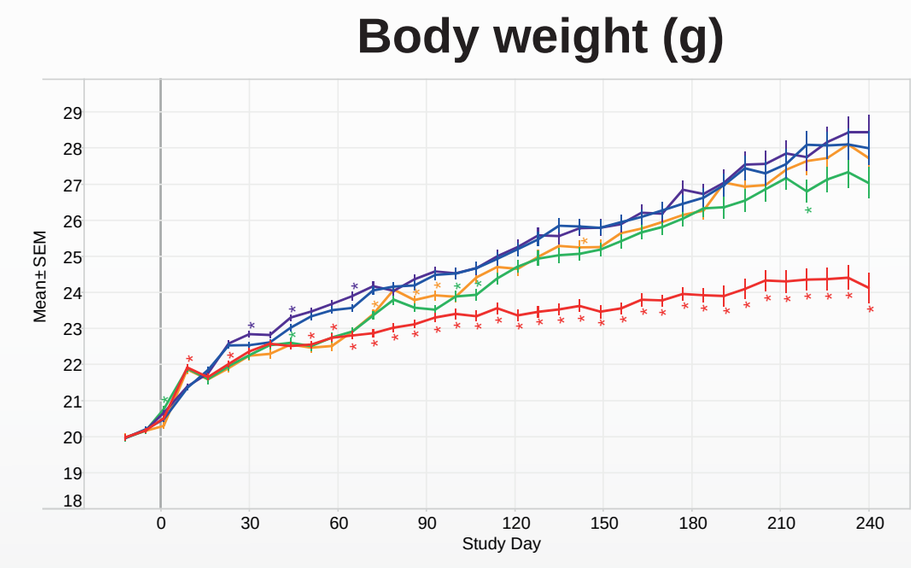
Body weight (g)
Mean±SEM
Study Day

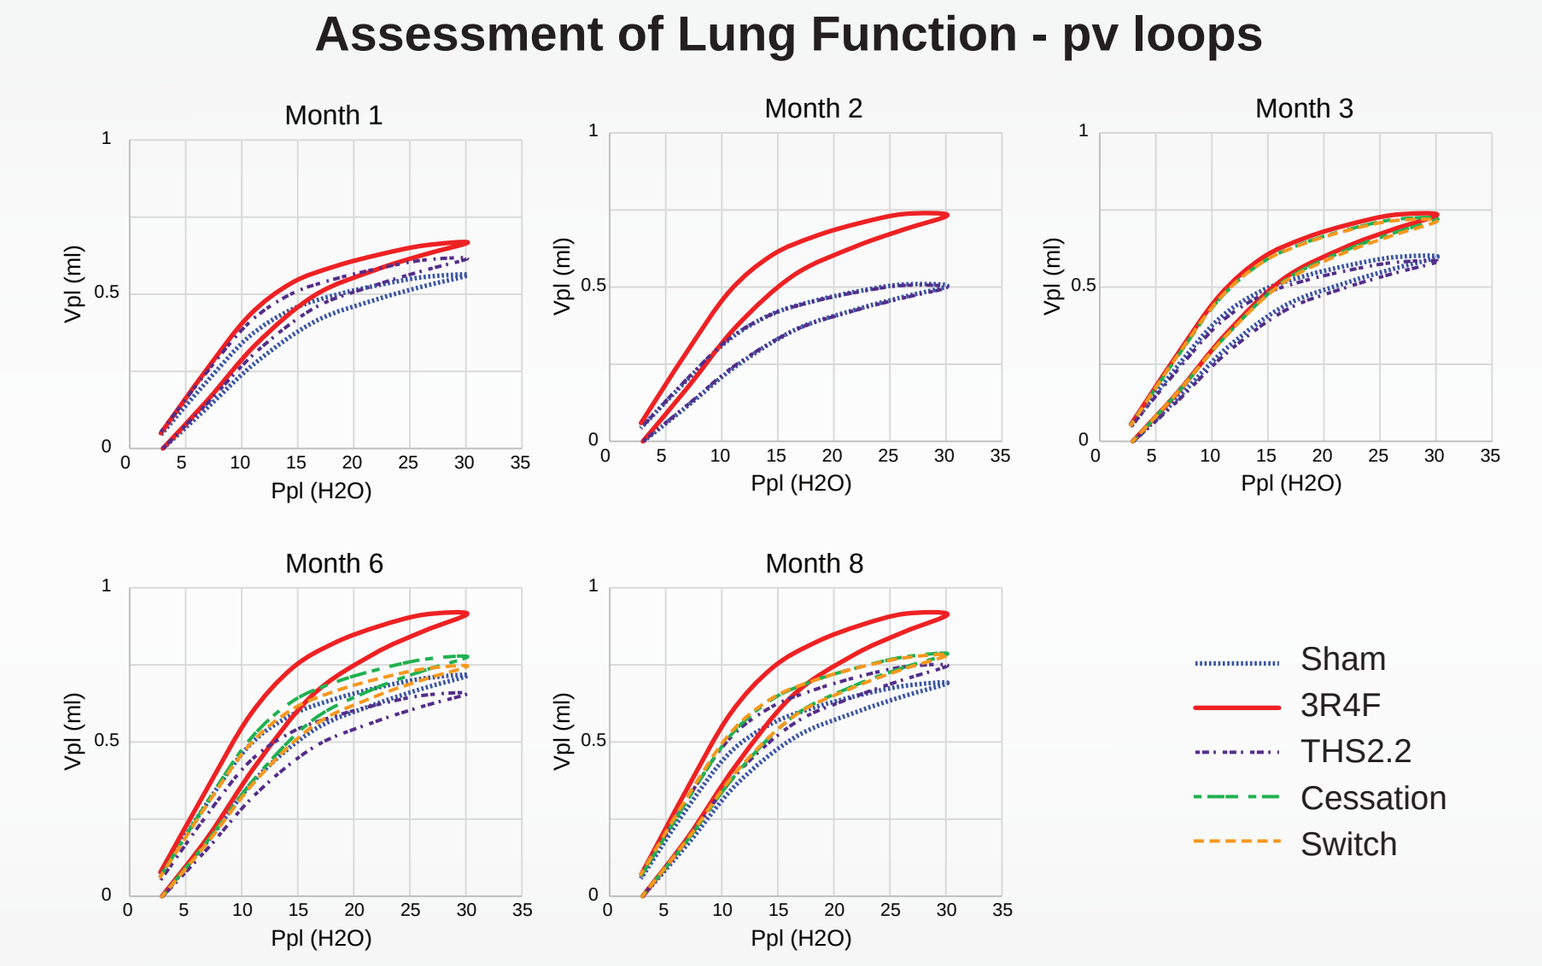
Assessment of Lung Function - pv loops
Month 1
Month 2
Month 3
Month 6
Month 8
Vpl (ml)
Ppl (H2O)
Sham
3R4F
THS2.2
Cessation
Switch

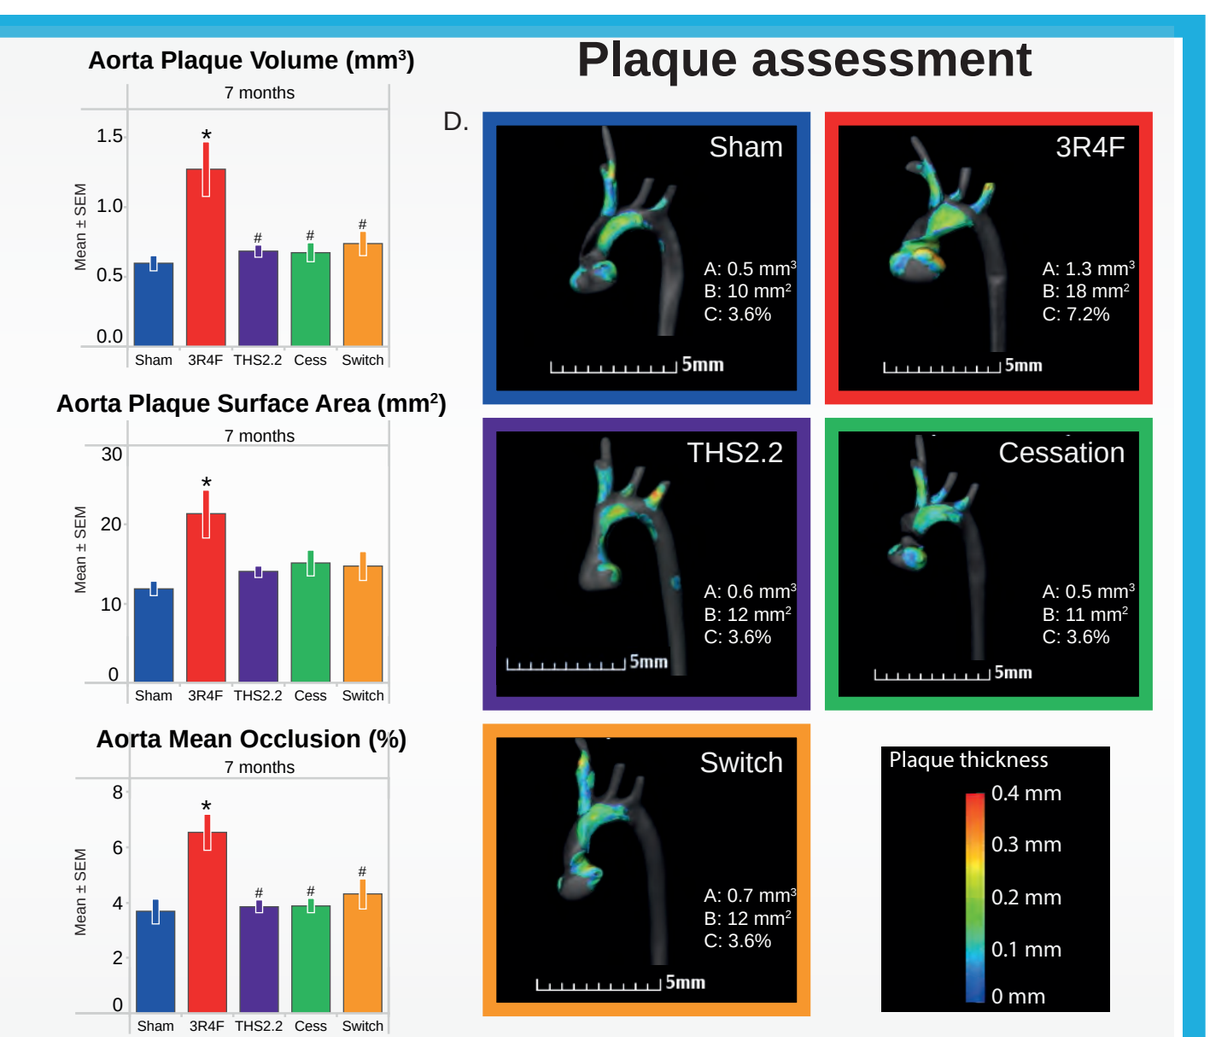
Aorta Plaque Volume (mm³)
Aorta Plaque Surface Area (mm²)
Aorta Mean Occlusion (%)
7 months
Plaque assessment
Sham
3R4F
THS2.2
Cessation
Switch
Plaque thickness

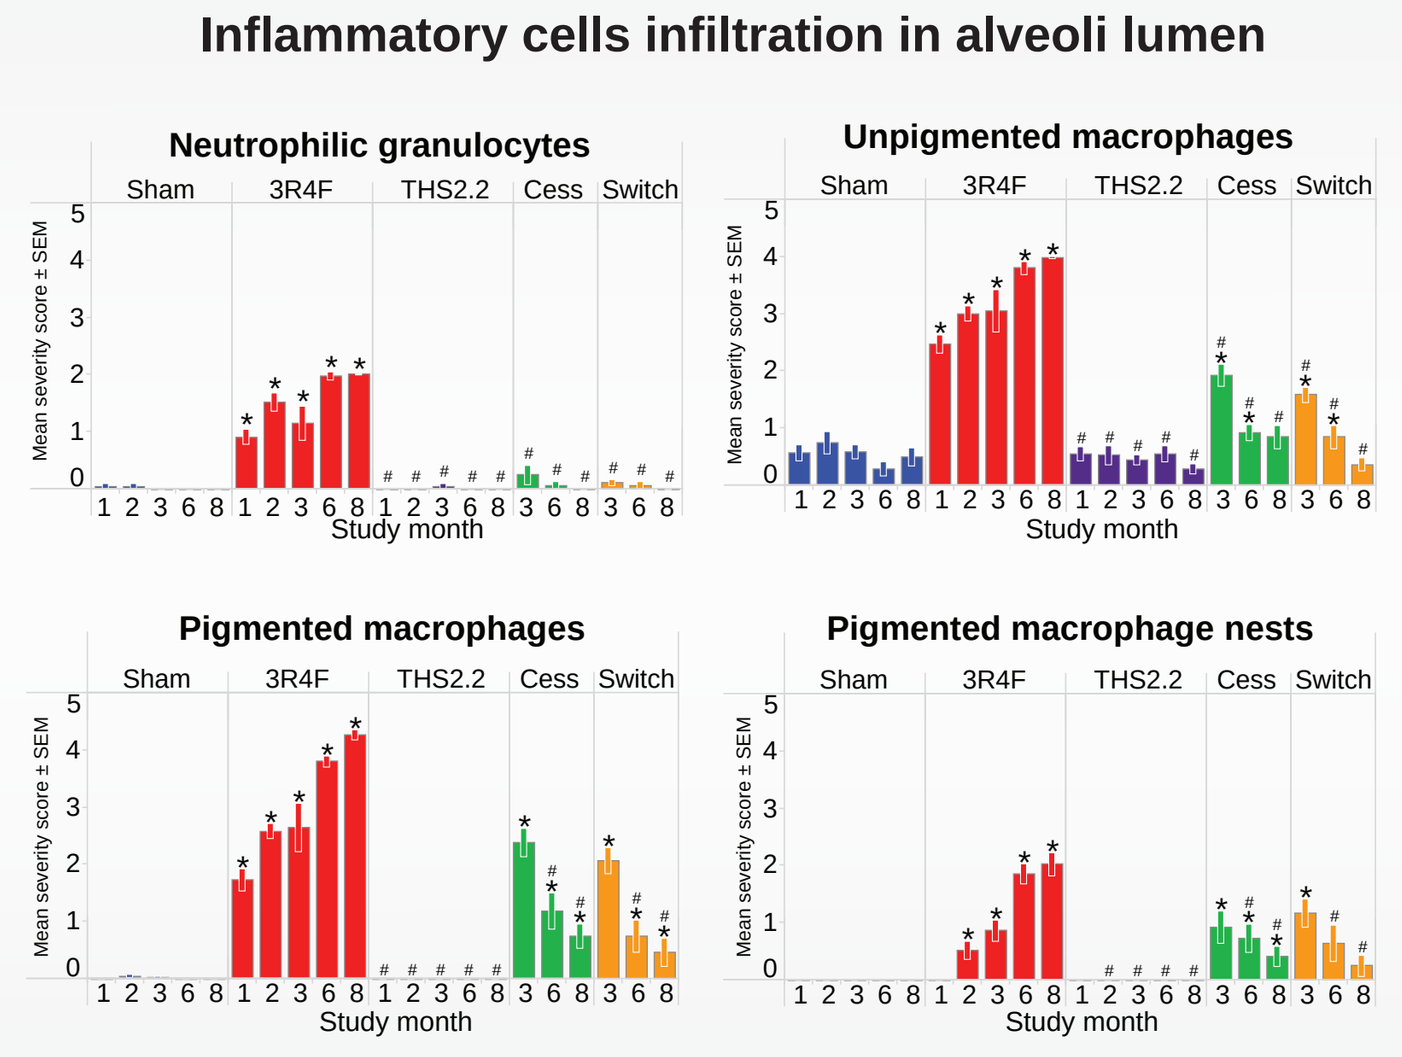
Inflammatory cells infiltration in alveoli lumen
Neutrophilic granulocytes
Unpigmented macrophages
Pigmented macrophages
Pigmented macrophage nests
Study month

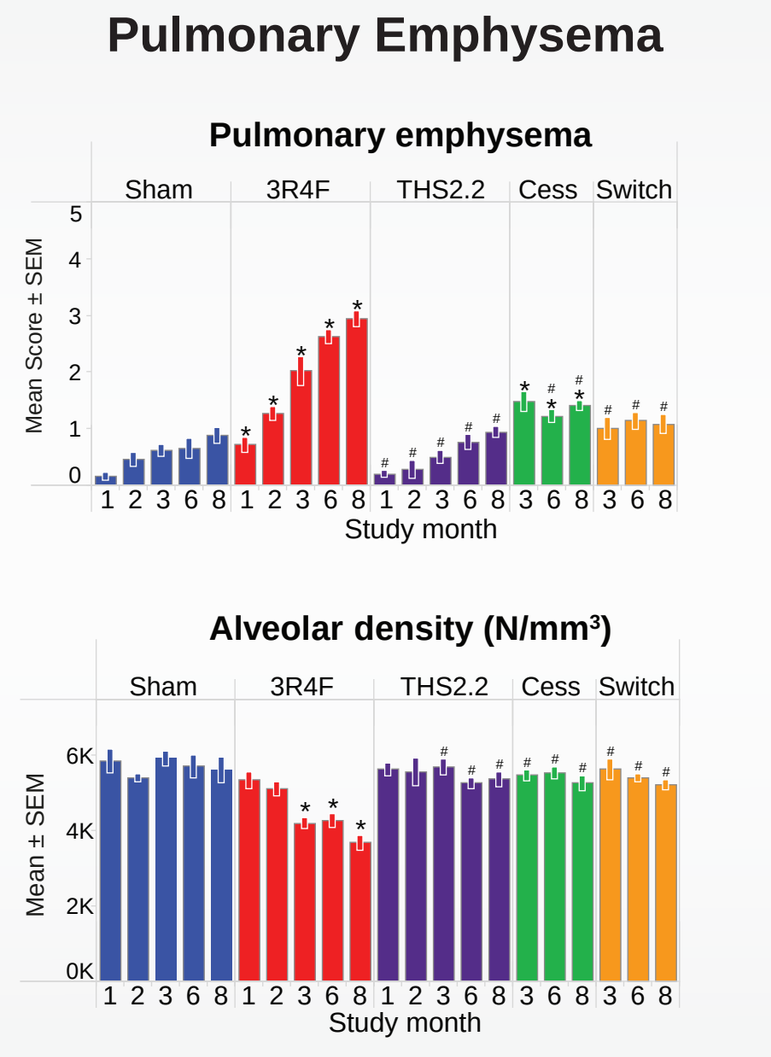
Pulmonary Emphysema
Pulmonary emphysema
Alveolar density (N/mm³)
Study month

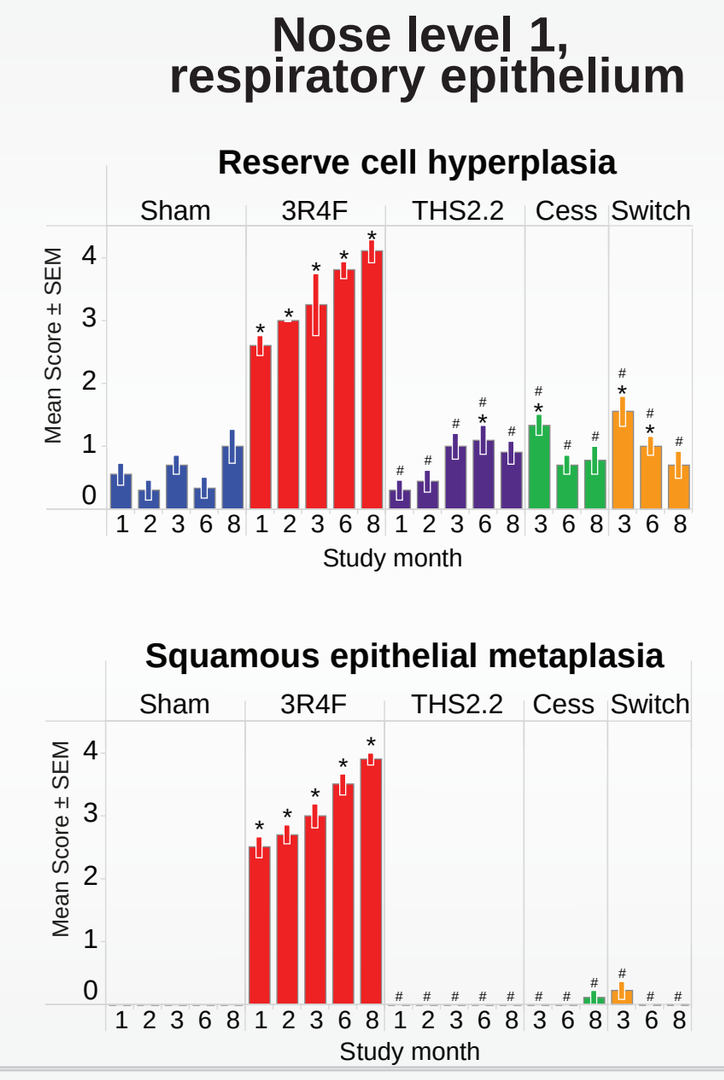
Nose level 1, respiratory epithelium
Reserve cell hyperplasia
Squamous epithelial metaplasia
Study month

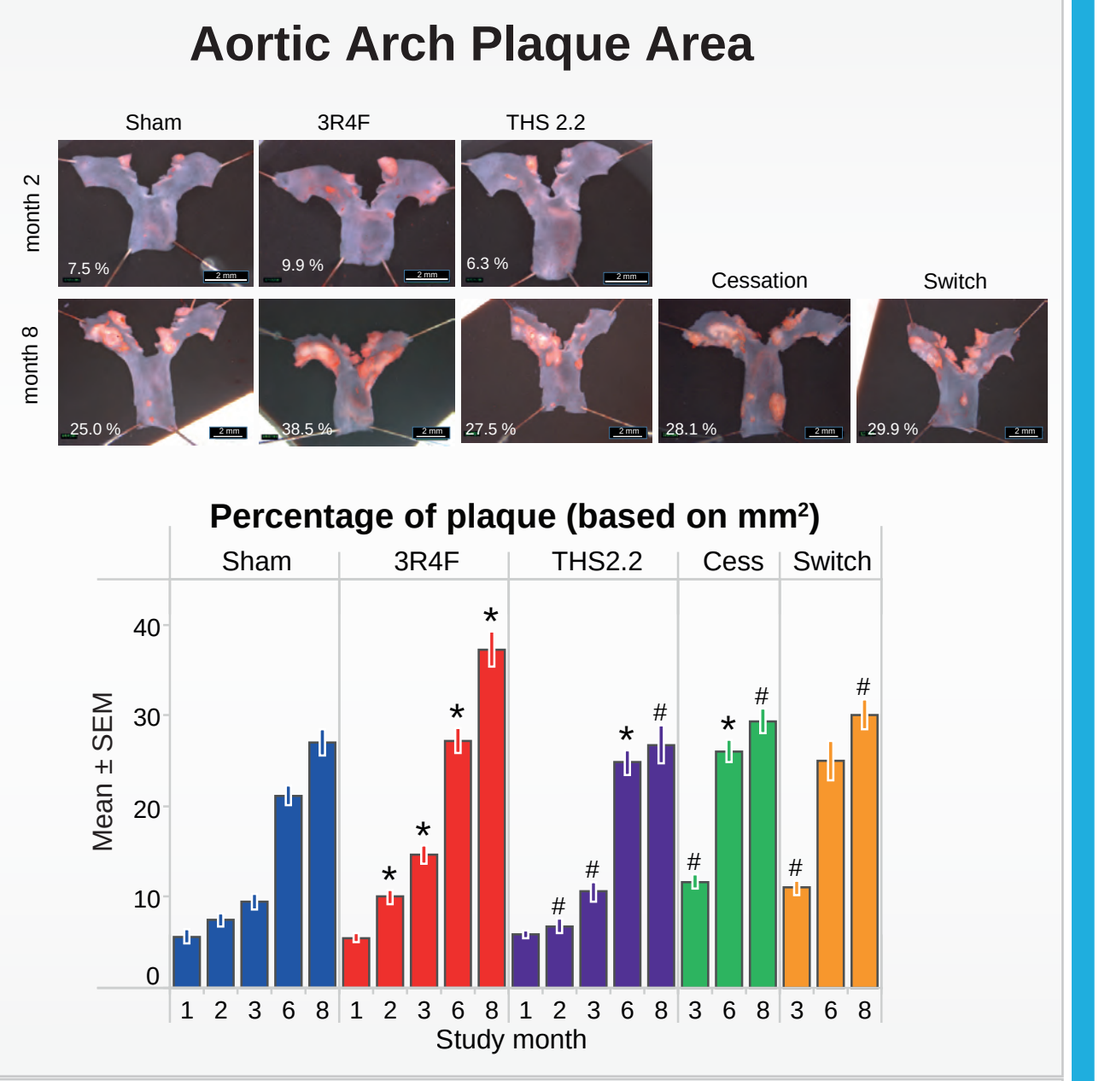
Aortic Arch Plaque Area
Sham
3R4F
THS 2.2
Cessation
Switch
month 2
month 8
Percentage of plaque (based on mm²)
Study month

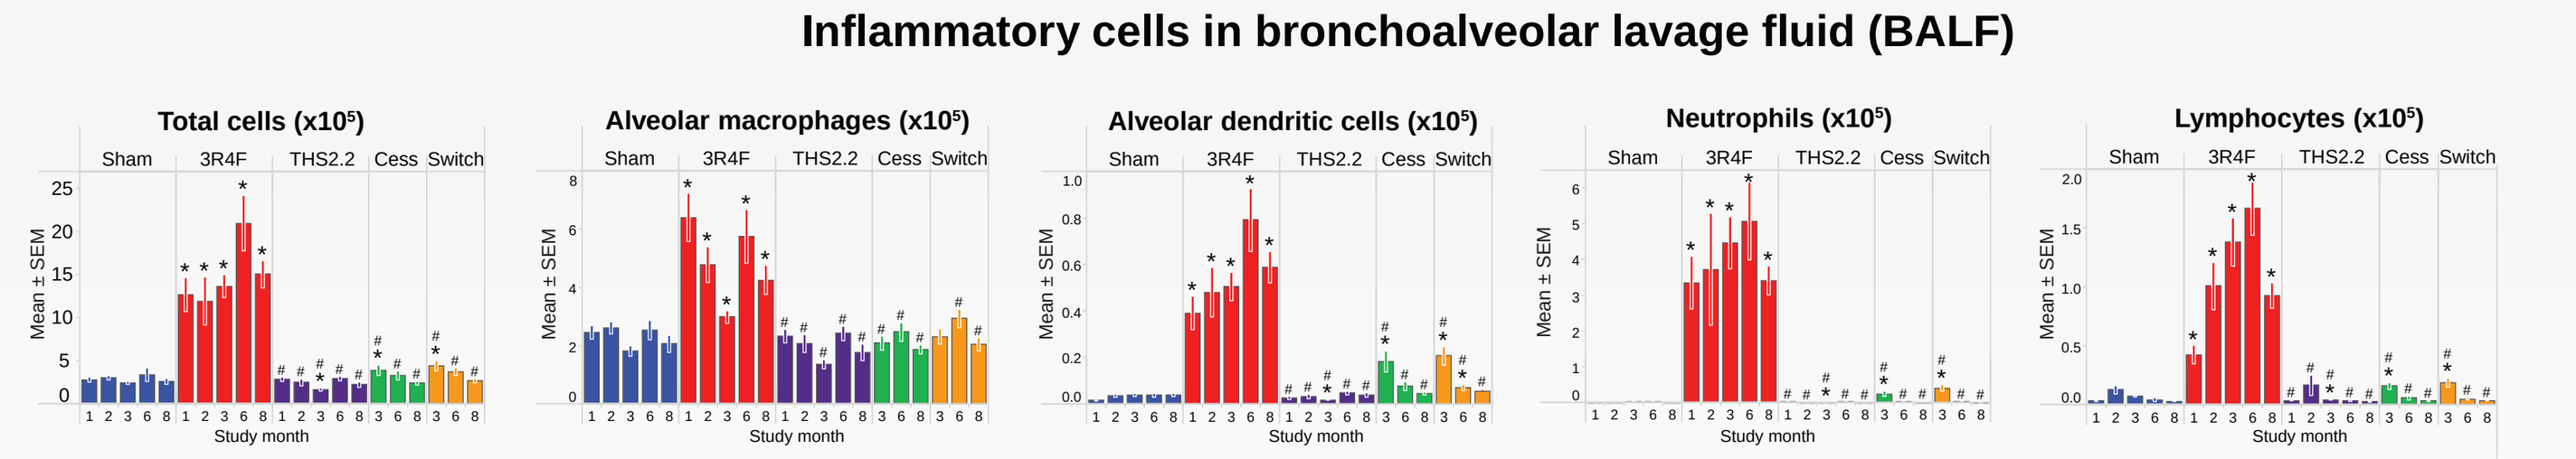
Inflammatory cells in bronchoalveolar lavage fluid (BALF)
Total cells (x10⁵)
Alveolar macrophages (x10⁵)
Alveolar dendritic cells (x10⁵)
Neutrophils (x10⁵)
Lymphocytes (x10⁵)
Study month

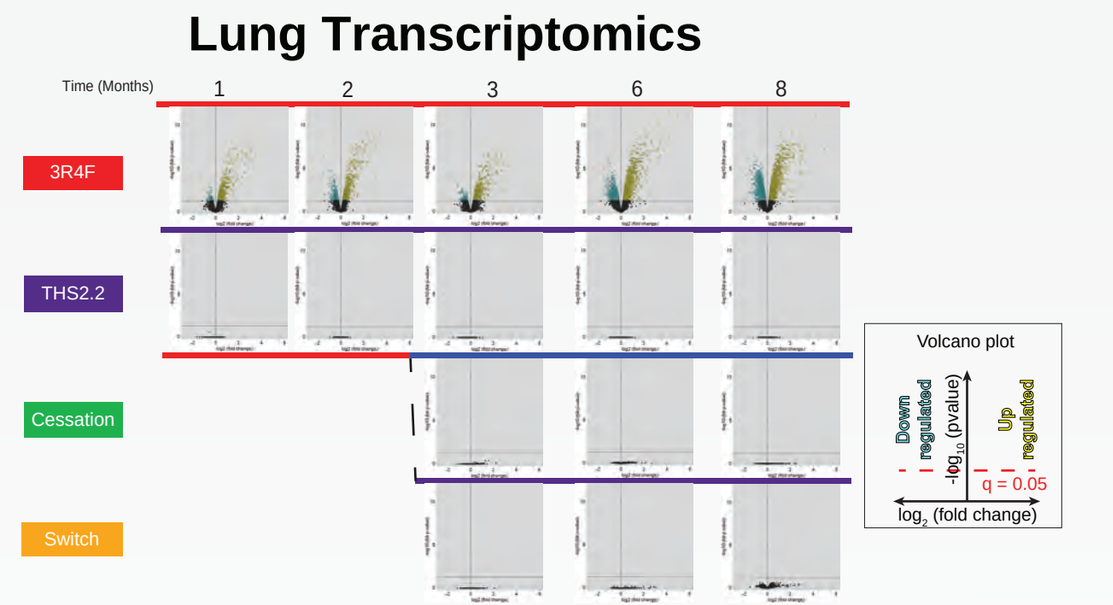
Lung Transcriptomics
3R4F
THS2.2
Cessation
Switch
Volcano plot
q = 0.05
log₂ (fold change)

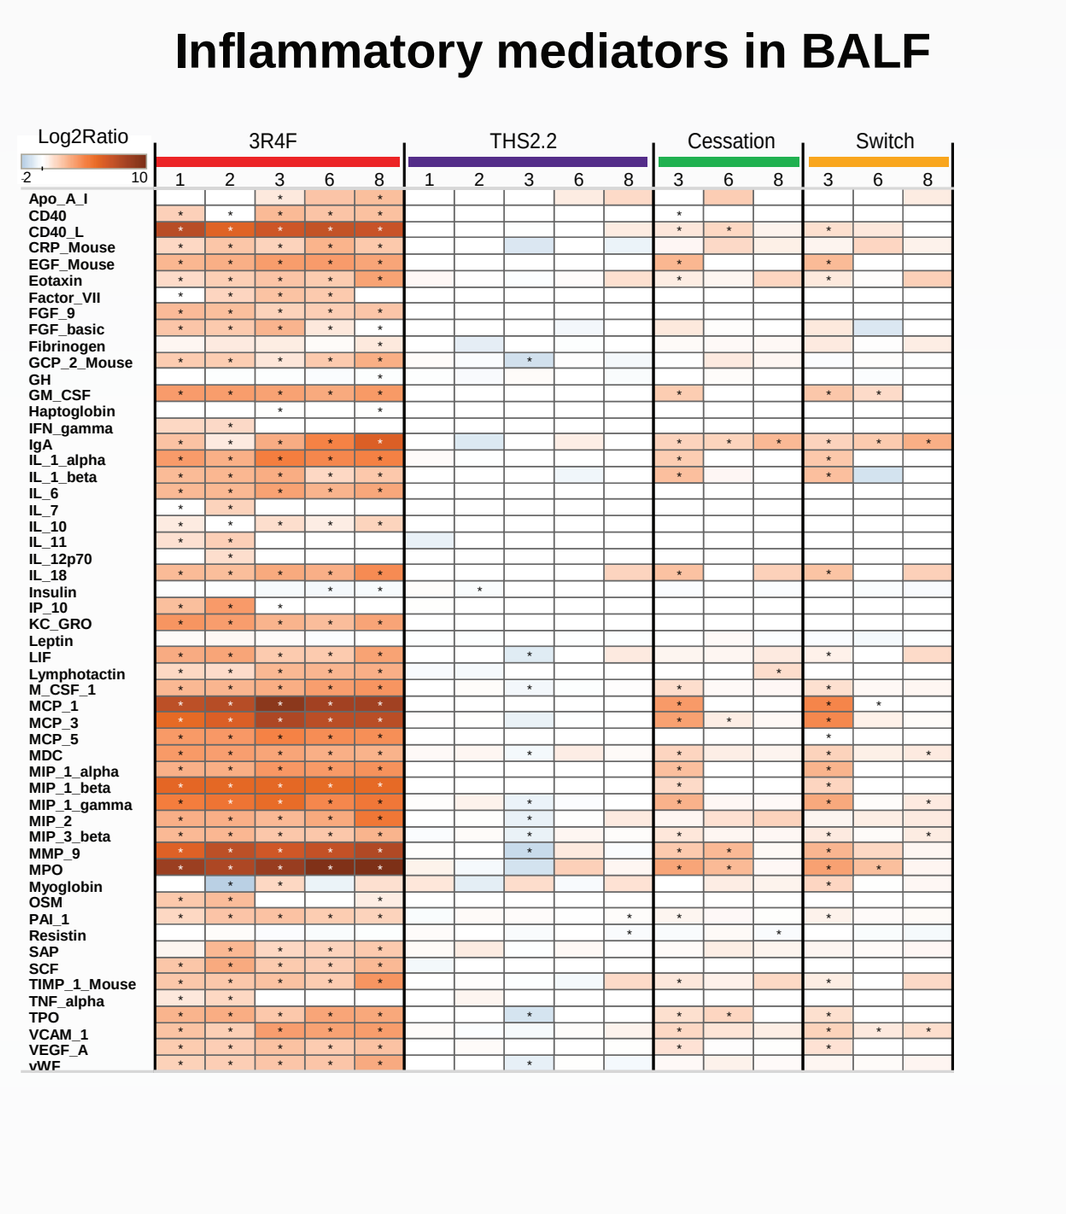
Inflammatory mediators in BALF
Log2Ratio
3R4F
THS2.2
Cessation
Switch

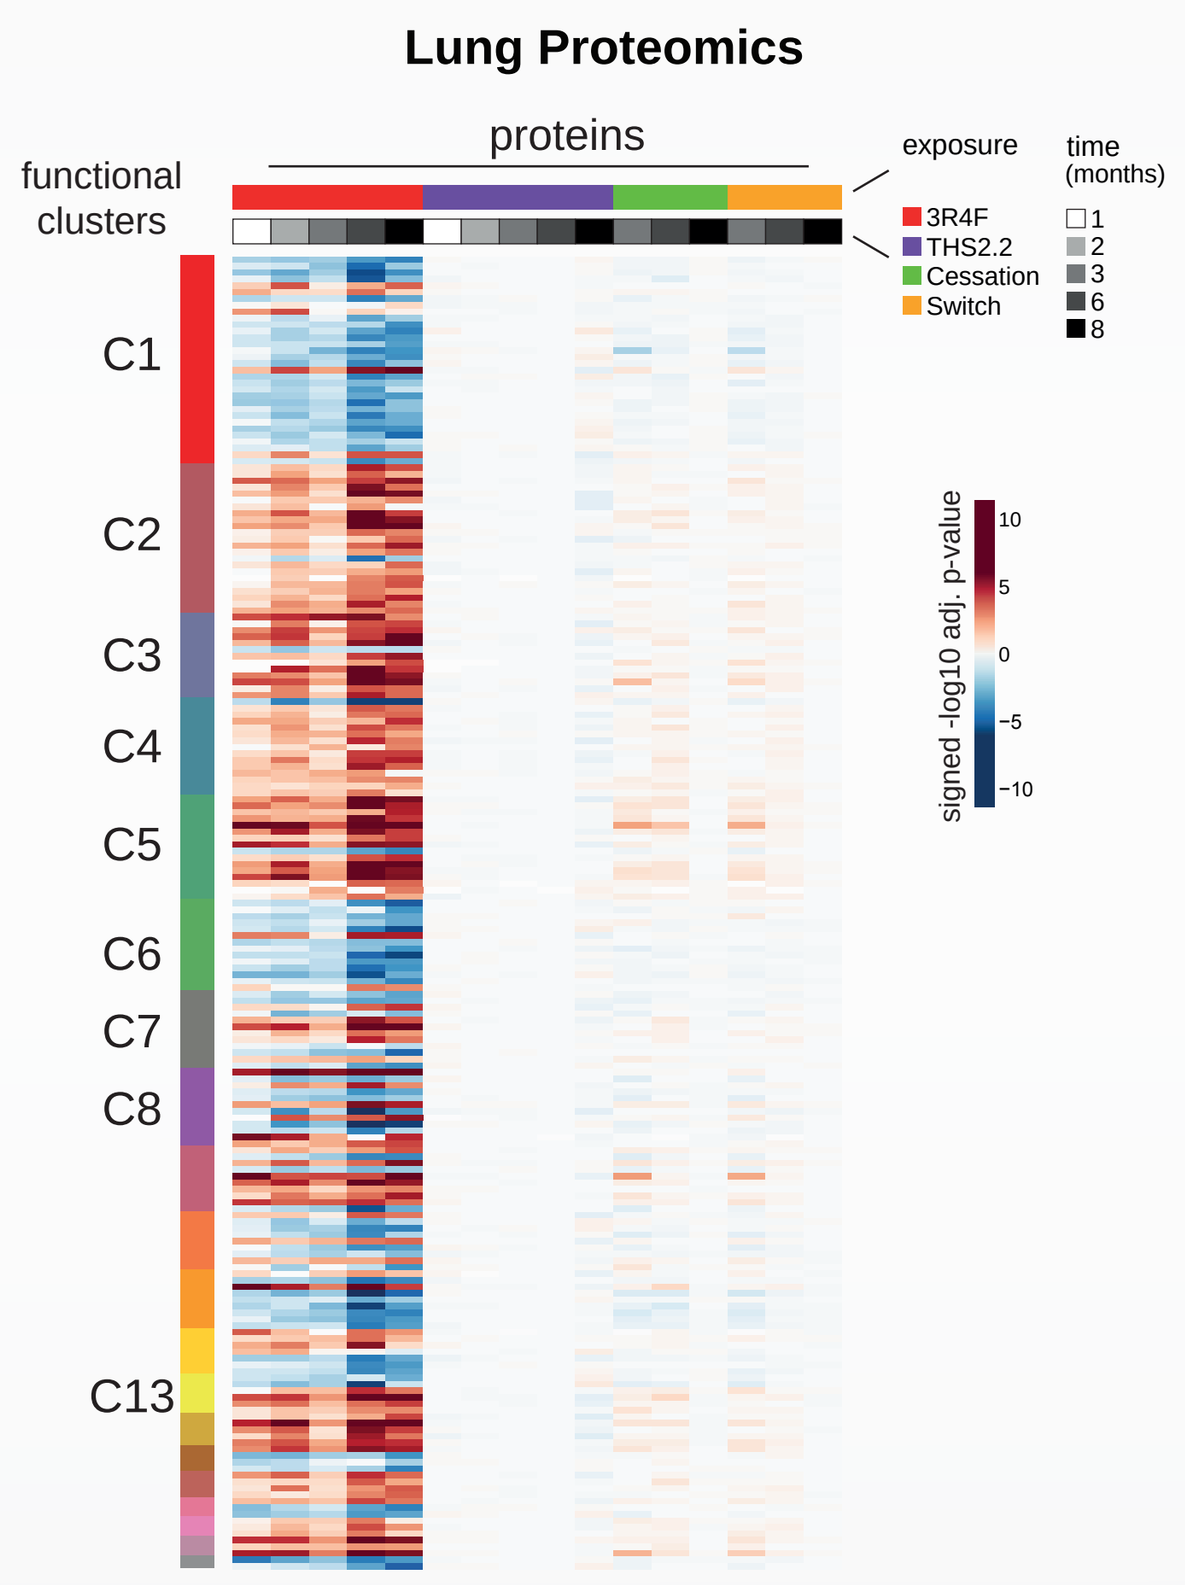
Lung Proteomics
proteins
functional clusters
exposure
time (months)
3R4F
THS2.2
Cessation
Switch
signed -log10 adj. p-value

C1: cell junction, C2: oxidative phosphorylation
C3: oxidative stress, C4: unfolded-protein response,
C5: lysosome, immune-related, C6: ECM & ECM interaction,
C7: metabolism, e.g. pentose-phosphate pathway,
C8: xenobiotic stress, and C13: surfactants

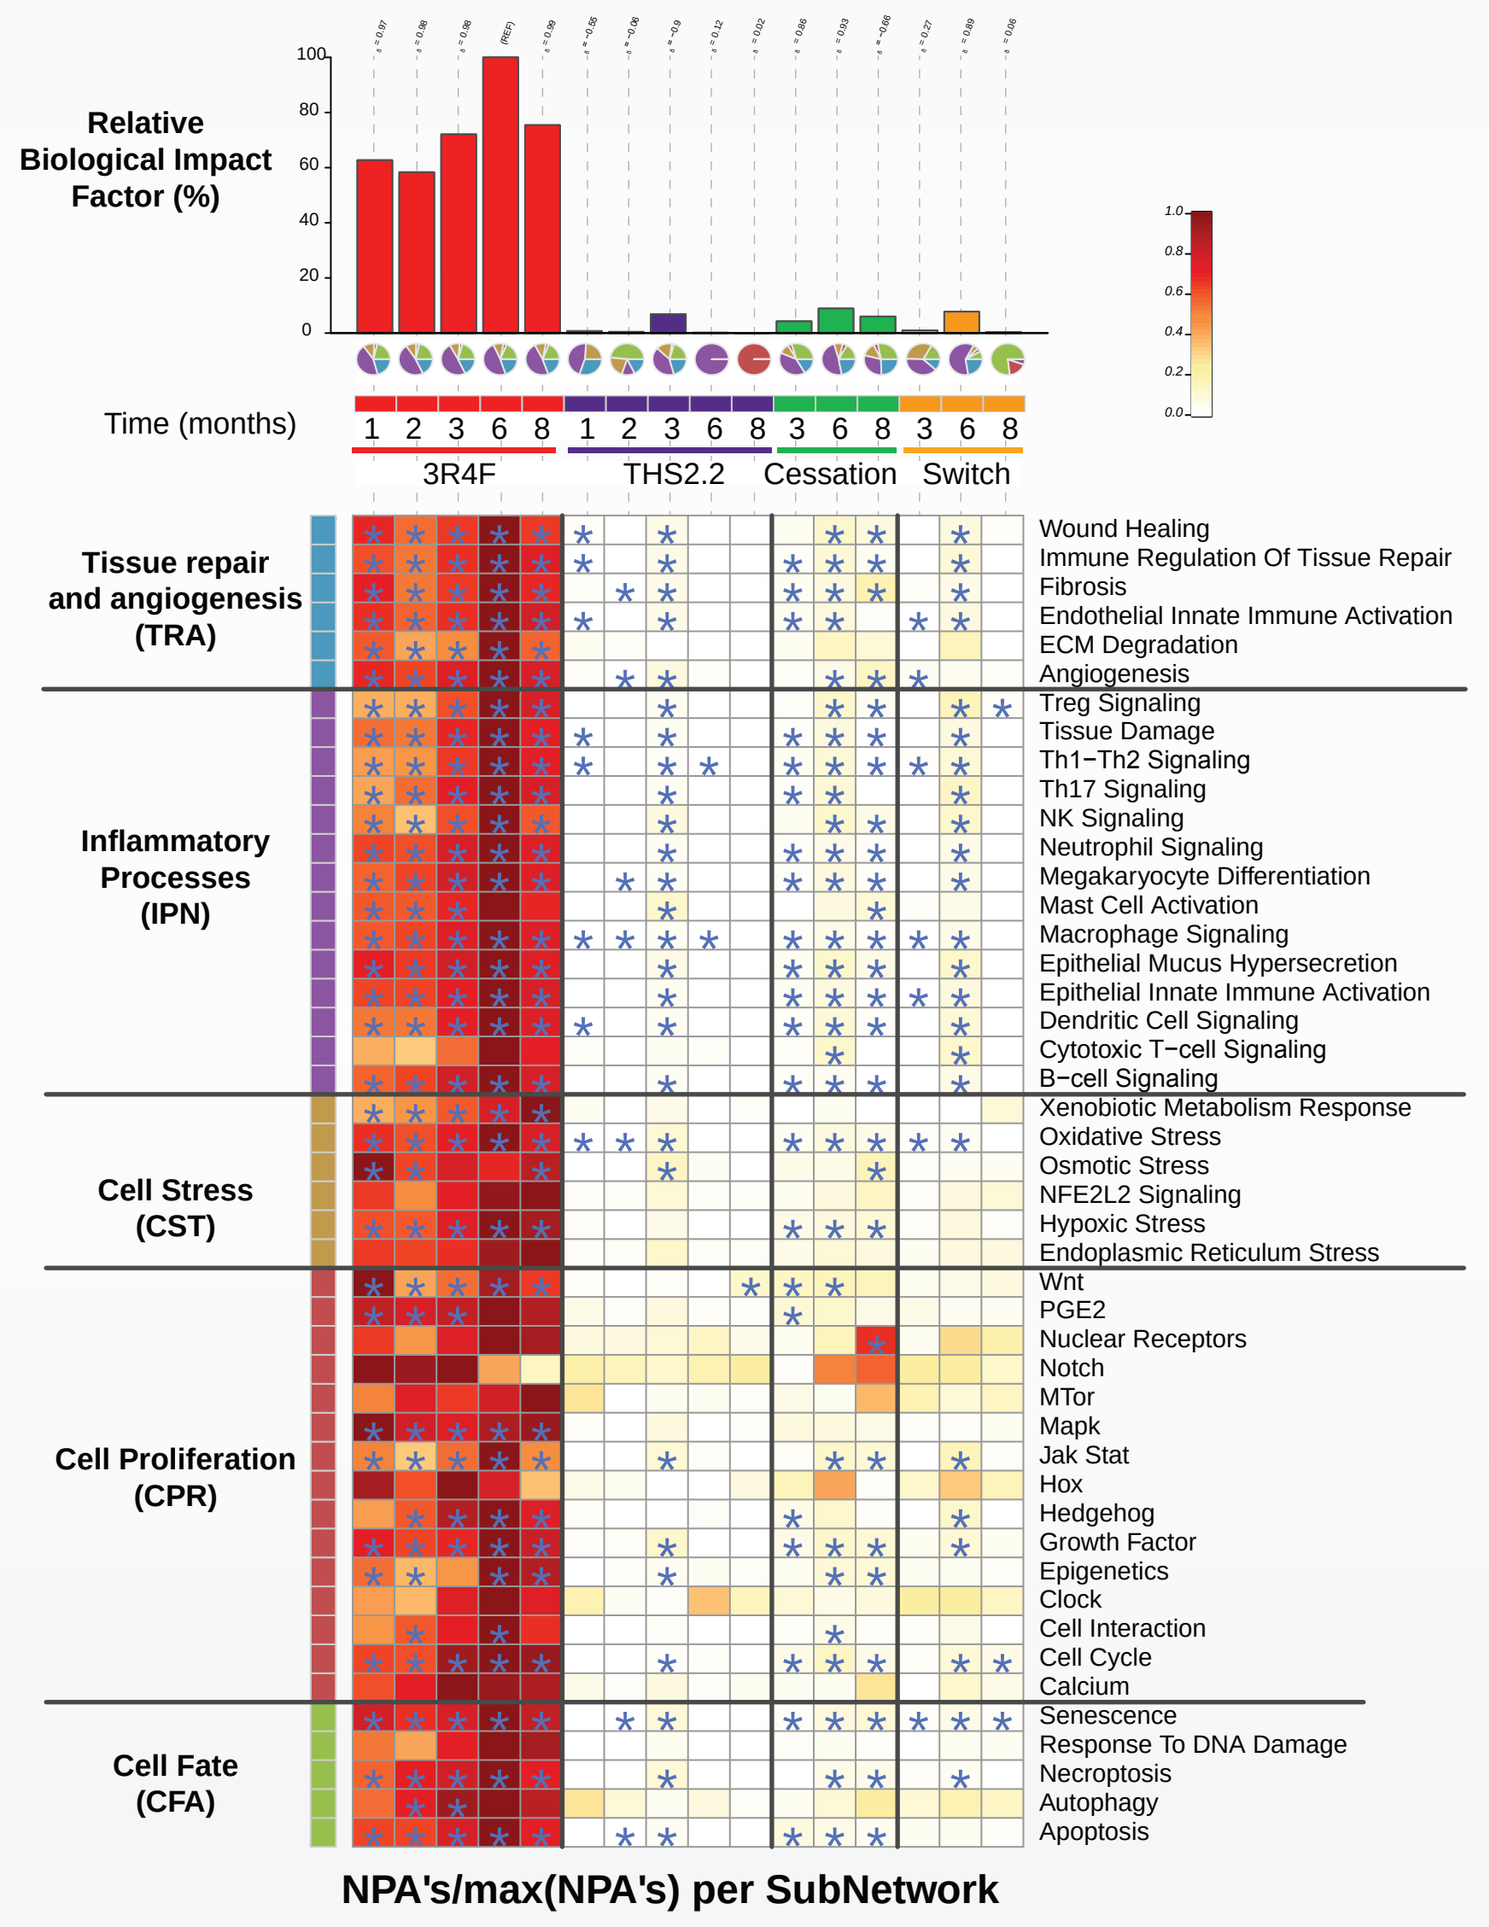
Relative Biological Impact Factor (%)
Time (months)
3R4F
THS2.2
Cessation
Switch
Tissue repair and angiogenesis (TRA)
Inflammatory Processes (IPN)
Cell Stress (CST)
Cell Proliferation (CPR)
Cell Fate (CFA)
NPA's/max(NPA's) per SubNetwork

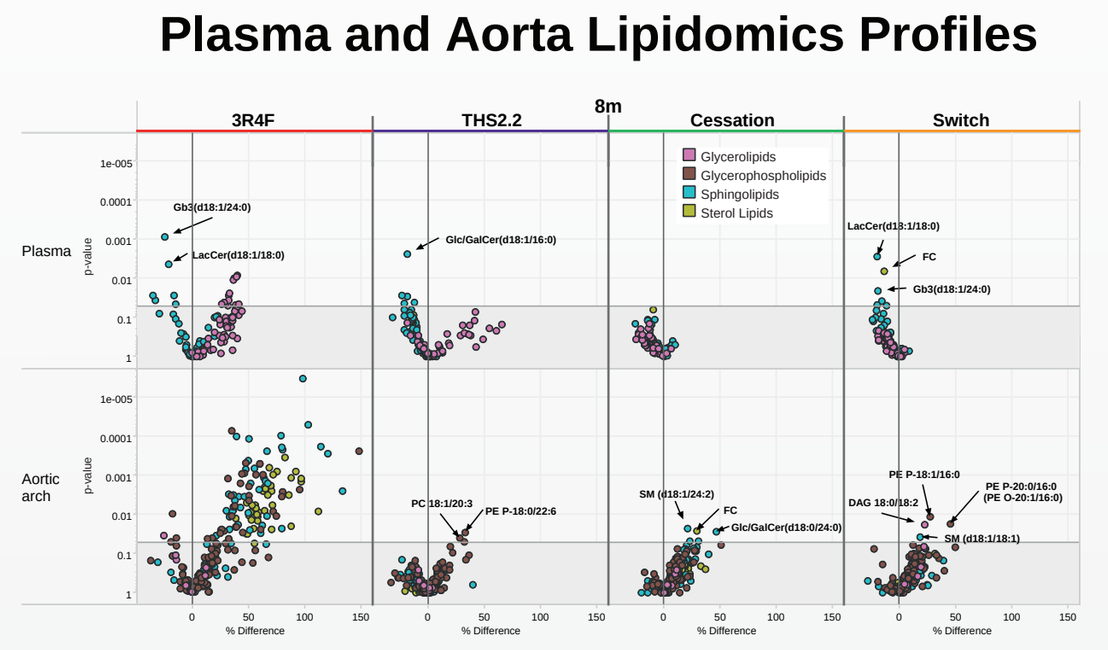
Plasma and Aorta Lipidomics Profiles
3R4F
THS2.2
Cessation
Switch
Plasma
Aortic arch

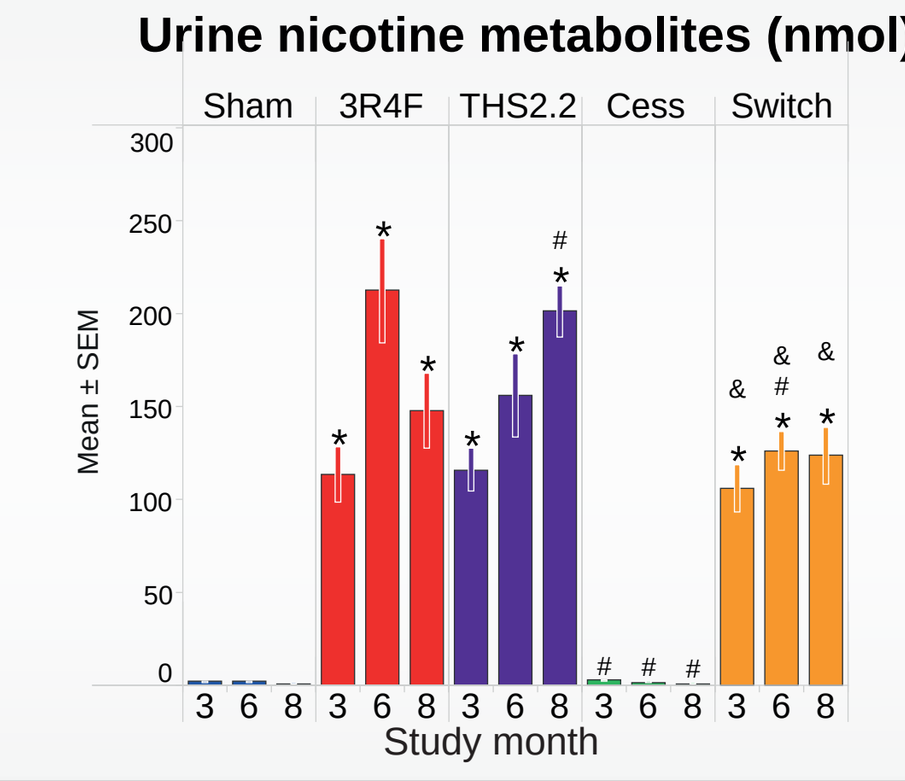
Urine nicotine metabolites (nmol)
Mean ± SEM
Study month

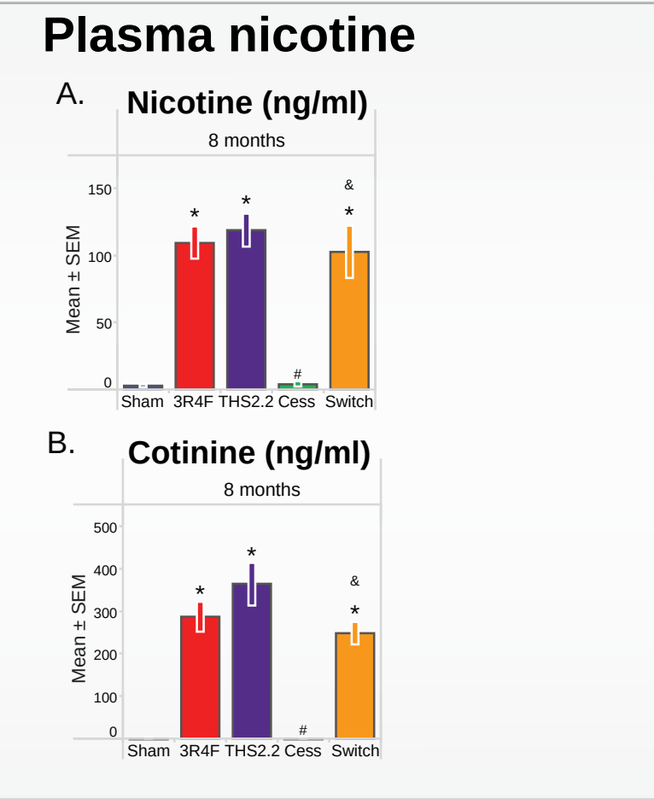
Plasma nicotine
Nicotine (ng/ml)
Cotinine (ng/ml)
8 months

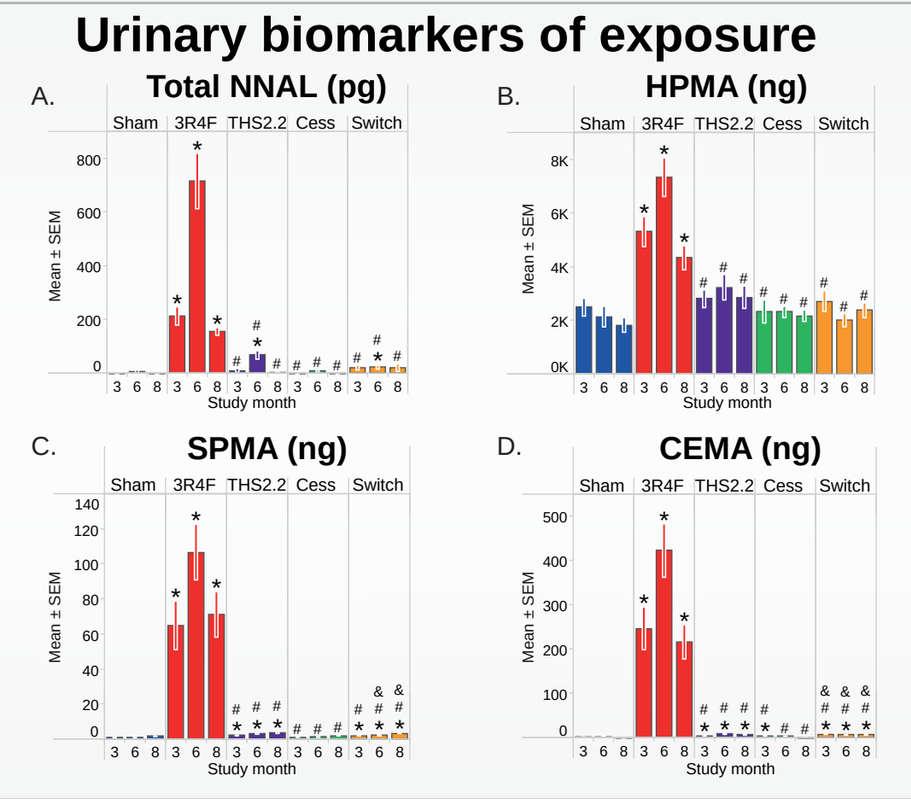
Urinary biomarkers of exposure
Total NNAL (pg)
HPMA (ng)
SPMA (ng)
CEMA (ng)

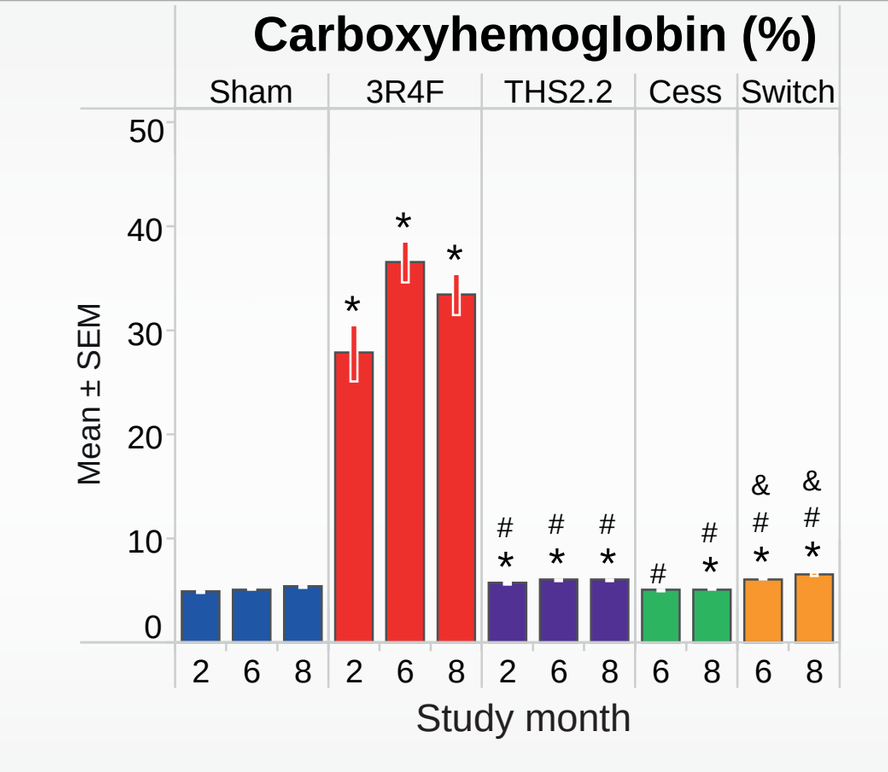
Carboxyhemoglobin (%)
Mean ± SEM
Study month

Experimental Design

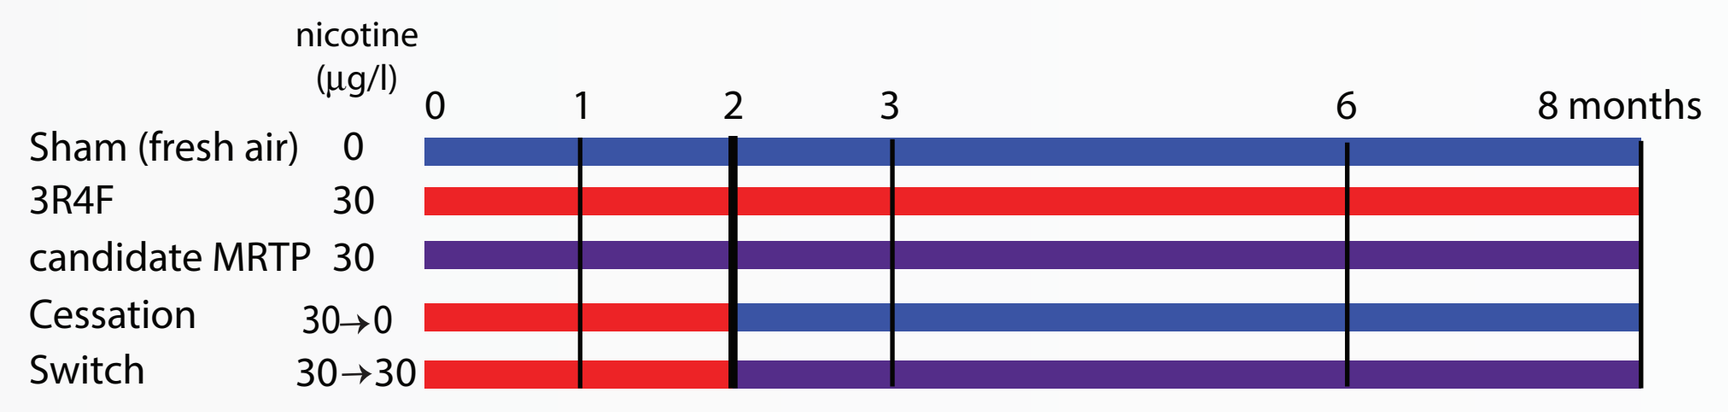
nicotine (µg/l)
0
1
2
3
6
8 months
Sham (fresh air) 0
3R4F 30
candidate MRTP 30
Cessation 30→0
Switch 30→30

Female Apoe$^{-/-}$ mice were exposed to 3R4F (600 mg/m^3 TPM), THS2.2 (matched to the nicotine in 3R4F – 30 µg/l) or filtered air for 3 hours per day, 5 days per week, for up to 8 months. After 2 months of exposure to 3R4F, switching and cessation groups were exposed to aerosol from THS2.2 or filtered air, respectively. Animals were observed on a daily basis, body weight progression was monitored twice per week, exposure parameters (carboxyhemoglobin (COHb) in blood and nicotine metabolites in urine) were measured 3 times during the study. Dissections were performed after 1, 2, 3, 6, and 8 months of exposure. At each time point animals were allocated for the following end points: bronchoalveolar lavage fluid (BALF), identification of infiltrated inflammatory cells in lungs and multi-analyte (cytokines/chemokines) profiling; histopathological evaluation and morphometry of lungs; lung function; plaque surface determination and an extensive molecular high throughput analysis (transcriptomics, proteomics and lipidomics).

Test Atmosphere Chemistry

	Sham	3R4F	THS2.2
TPM (mg/m³)	0 (250)	598.5 ± 27.1 (256)	368.9 ± 54.4 (250)
Nicotine (mg/m³)	0 (63)	29.4 ± 2.2 (256)	28.6 ± 3.2 (250)
Carbon monoxide (ppm)	0.1 ± 0.1 (250)	650.9 ± 34.2 (256)	14.5 ± 2.2 (250)
Acetaldehyde (mg/m³)	n.d.	32.9 ± 2.3 (50)	7.5 ± 0.7 (49)
Acrolein (mg/m³)	n.d.	3.1 ± 0.2 (50)	0.3 ± 0.0 (49)
Formaldehyde (mg/m³)	n.d.	0.6 ± 0.1 (50)	0.1 ± 0.0 (49)

Values given in the table are mean ± SD. Numbers in brackets are the number of measurements done during the study.

ACT 36th Annual Meeting
Red Rock Resort
Summerlin, Nevada
November 8–11, 2015

Philip Morris International Research & Development,
Quai Jeanrenaud 5, 2000 Neuchâtel, Switzerland
T: +41 58 242 21 13, F: +41 58 242 28 11, W: www.pmi.com