Reduced Impact of a Prototypic Modified Risk Tobacco Product Compared with Conventional Cigarette Smoke on Exposed Human and Rat Primary Normal Bronchial Epithelial Cells

P-1.104

Carole Mathis[1], Ulrike Kogel[1], Dimitris Messinis[2], Carine Poussin[1], Florian Martin[1], Stefan Frentzel[1], Emmanuel Guedj[1], Nikolai V. Ivanov[1], Julia Hoeng[1], Manuel C. Peitsch[1]

[1] Philip Morris International R&D,CH- 2000 Neuchâtel, Switzerland; [2] Protatonce Ltd, Scientific Park Lefkippos, Attiki, Greece

Introduction

Towards the twenty-first century new toxicity testing strategy, we have developed a systems biology-based approach[1] that integrates advanced, large-scale molecular measurements and cellular endpoints with computational methods to analyze and quantify biological network perturbations after exposure of toxicants such as cigarette smoke (CS).
We exposed for 4h two primary bronchial epithelial cell cultures from human and rat origin to different fractions (sbPBS, GVP, TPM) derived from reference CS (3R4F) or from a prototypic modified risk tobacco product (pMRTP) aerosol and analyzed their respective gene expression profiles compared to the corresponding control vehicles condition.

Using our systems toxicology-based assessment approach, we performed various comparative analysis:

- Between smoke fractions
- Between different doses of a same smoke fraction
- Between fractions from different tobacco products
- Across species

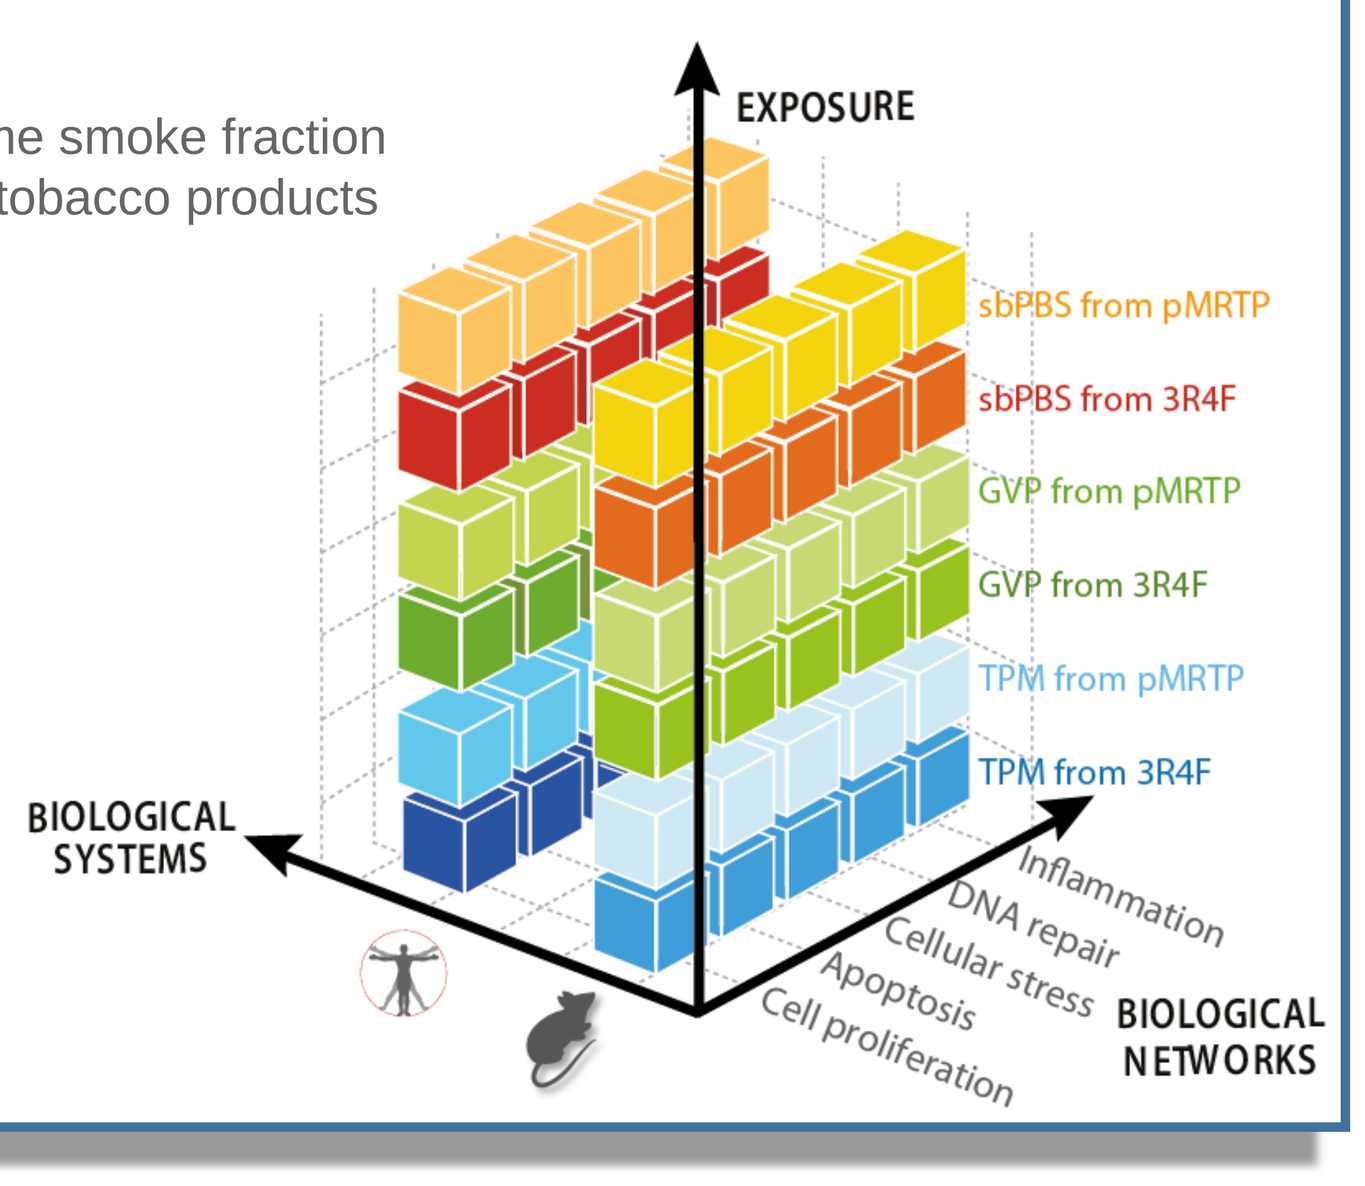
EXPOSURE
sbPBS from pMRTP
sbPBS from 3R4F
GVP from pMRTP
GVP from 3R4F
TPM from pMRTP
TPM from 3R4F
BIOLOGICAL SYSTEMS
Inflammation
DNA repair
Cellular stress
Apoptosis
Cell proliferation
BIOLOGICAL NETWORKS

Figure 1: This cubic experimental design space recapitulates the two biological systems (namely Normal Human Bronchial Epithelial cells (NHBE) and Normal Rat Bronchial Epithelial (NRBE) cells), the exposure regimen (GVP, TPM and sbPBS derived from 3R4F and pMRTP smoke) and the biological networks used to assess the biological impact of the exposure presented within this study.

Materials and Methods

Smoke fractions generation

Smoke from the reference cigarette 3R4F (from the University of Kentucky) and pMRTP sticks was generated under Health Canada Intense smoking regimen. Smoke bubbled-phosphate-buffered saline (sbPBS) was generated to trap water-soluble smoke fraction. Total Particulate Matter (TPM) of the mainstream smoke from 3R4F or pMRTP was trapped on a Cambridge filter before being extracted in ethanol. Gas Vapor Phase (GVP) corresponds to the remaining smoke from 3R4F or pMRTP bubbled in PBS after TPM trapping on filter. Fresh fractions were then diluted in culture medium just before exposure.

Cell culture

NHBE cells, purchased from Lonza Inc. (Switzerland) were derived from tracheo/bronchial epithelial tissue of a 60 years old male donor, without smoking history. NRBE cells (from CHI Scientific Inc., USA) were isolated from pooled tracheobronchial tissue of adult inbred AGA rats.

Exposure

Both NHBE and NRBE cells were cultured under the same condition and exposed in parallel to the same fraction of both test item and to control vehicles (PBS or ethanol) for 4h before RNA extraction.
The following doses were tested:

- For NHBE cells:
 - 24 puffs/L
 - 100 puffs/L
- For NRBE cells:
 - 6 puffs/L
 - 24 puffs/L

GVP / TPM / sbPBS — 3R4F / pMRTP

Gene expression data

Total RNA samples from NHBE and NRBE cells were hybridized to GeneChip Human Genome U133 Plus 2.0 Array (54'675 probesets) and GeneChip Rat Genome 203 2.0 Array (31'099 probesets), respectively. Human specific and rodent (mouse and rat) specific networks have been used to quantify the perturbations measured in NHBE and NRBE cells, respectively.

Figure 2: (A) The causal biological networks are describing biological processes or mechanisms (e.g., Cell Proliferation[2], Cell Stress[3], DNA damage and Apoptosis[5] or Inflammation[6]). They are composed of backbone nodes (big grey balls) connected by causal directional relationships (= edges) derived from an evidence line extracted from literature. Differential expression of genes (small black balls) are experimental evidences for the activation of upstream backbone node. (B) System response profiles (=differentially expressed genes) are translated into Network Perturbation Amplitude (NPA) scores[4] for each biological networks and sub-networks allowing a higher granularity of the biological interpretation of the dataset.
The Biological Impact Factor (BIF)[7] is computed by aggregating NPA scores. It represents a hollistic score that describes the system-wide effect of all biological processes perturbed after exposure.

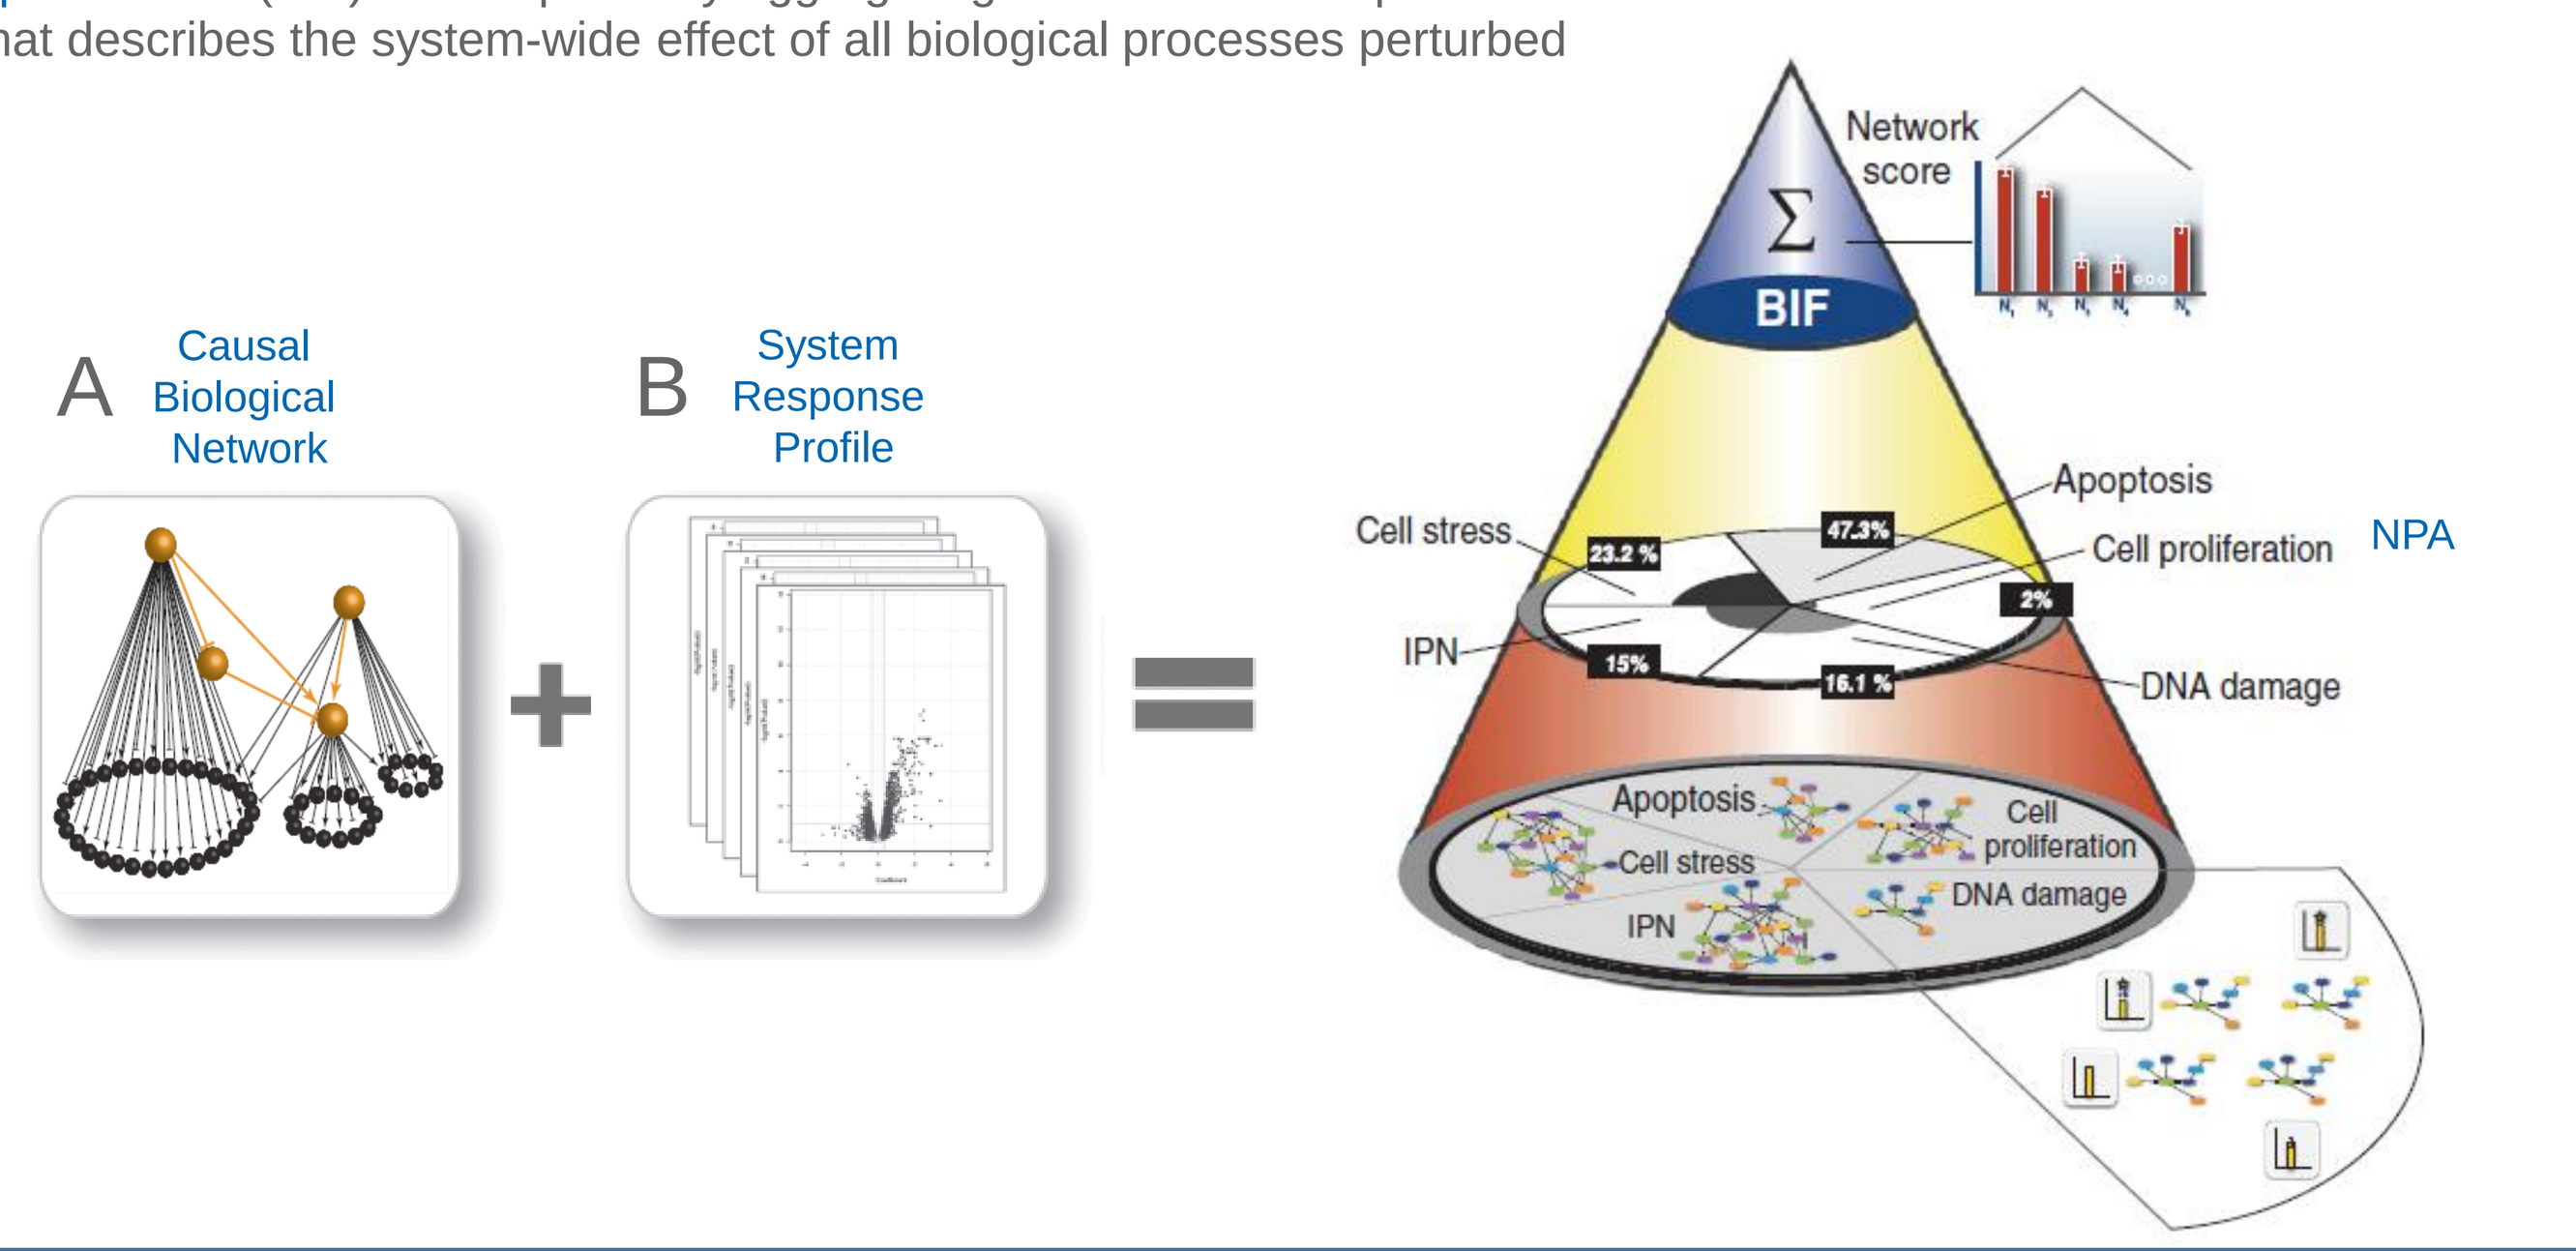
A Causal Biological Network
B System Response Profile
Network score
BIF
Apoptosis
Cell proliferation
NPA
Cell stress
IPN
DNA damage
47.3%
23.2 %
2%
15%
16.1 %

Results

Dose- and smoke fraction-dependent effect after 3R4F and pMRTP exposure in both human and rat bronchial epithelial cells

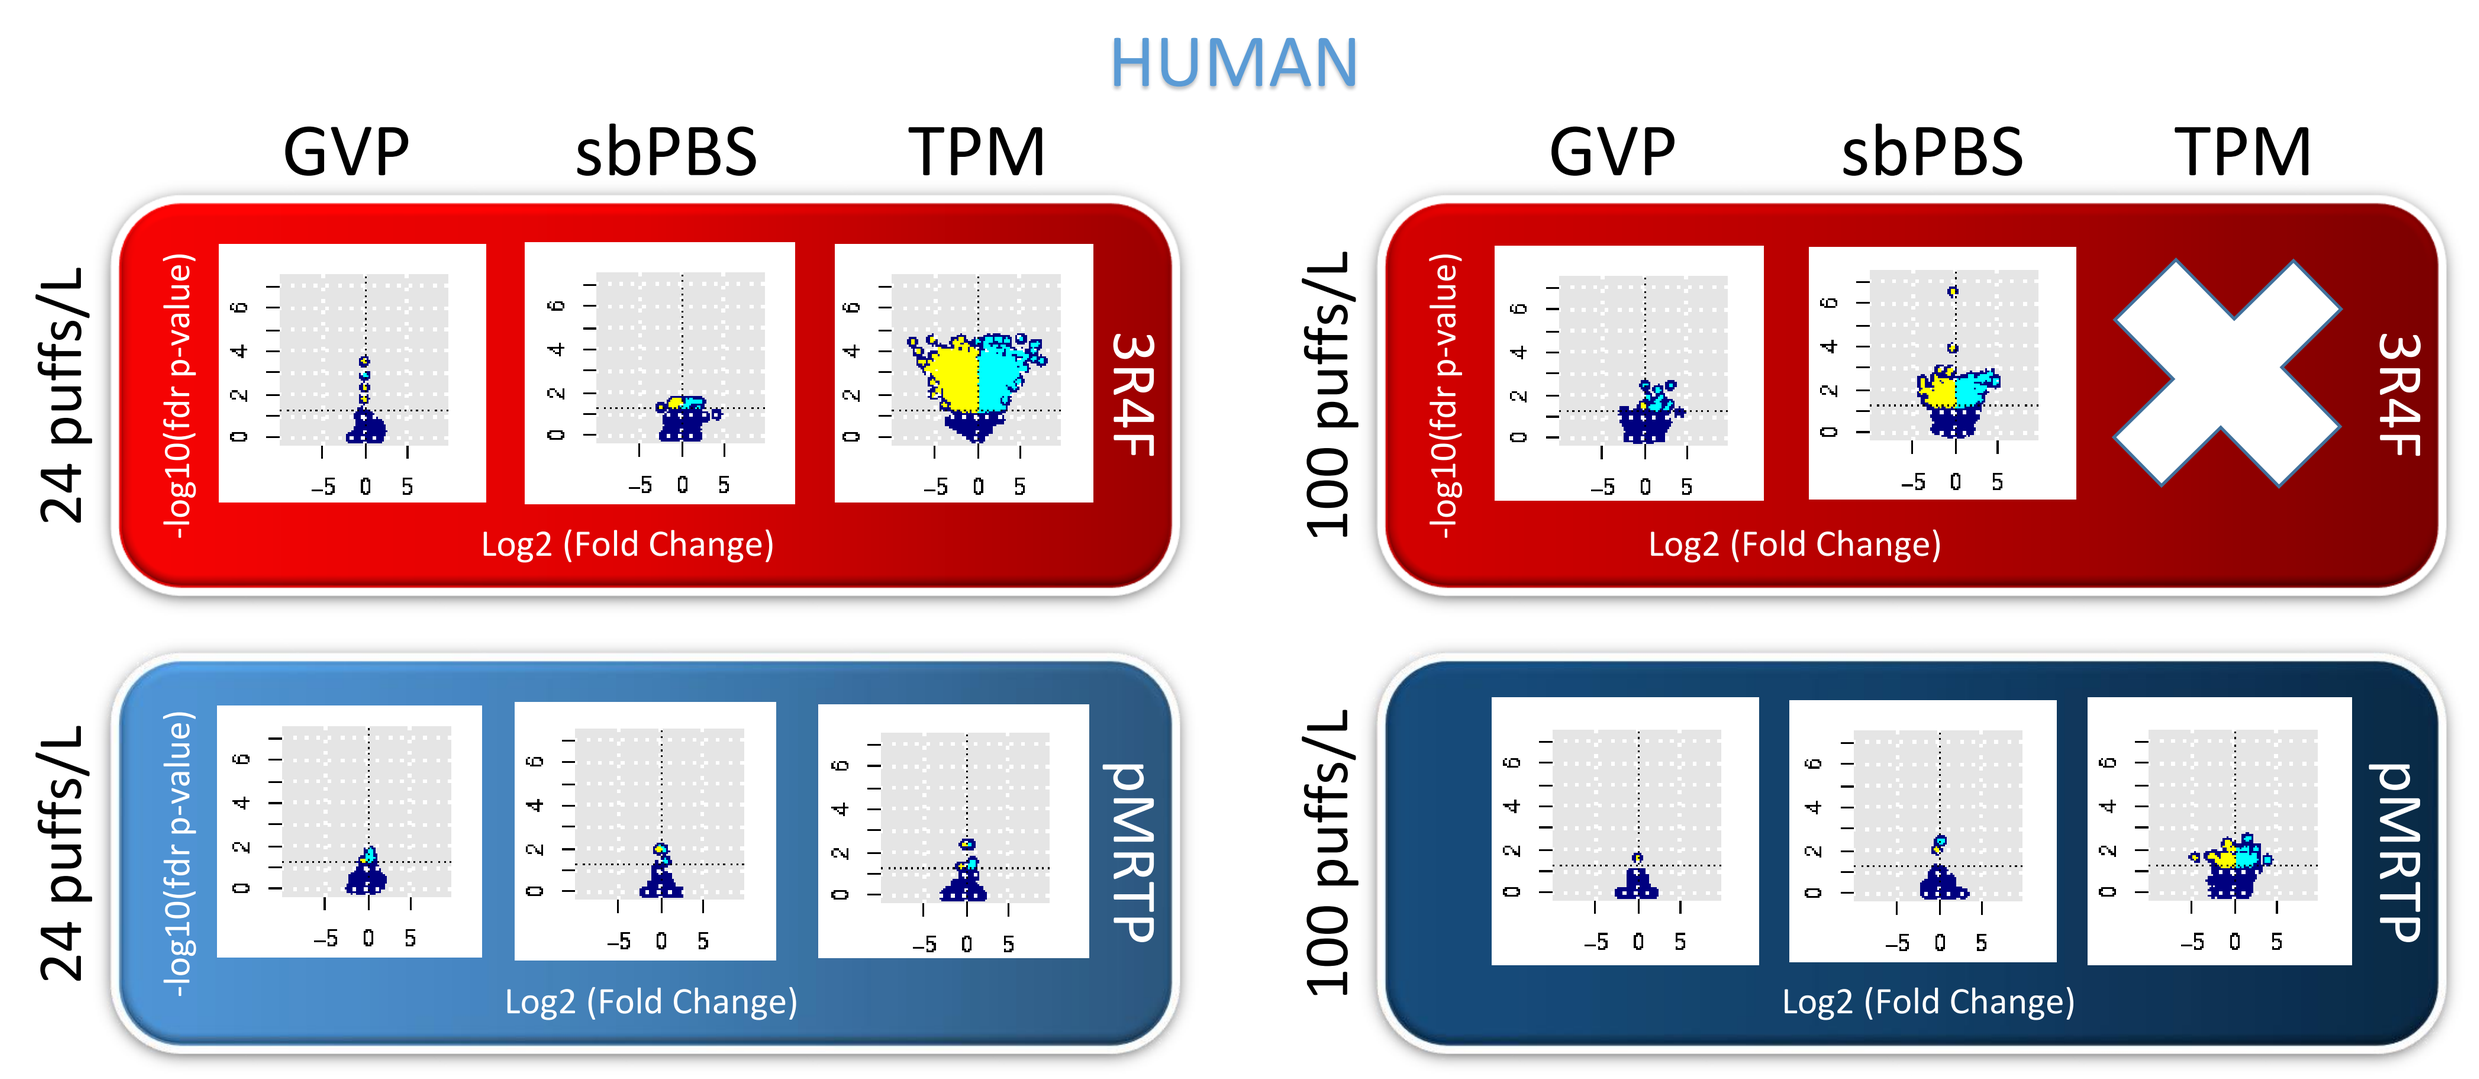
HUMAN
GVP
sbPBS
TPM
24 puffs/L
100 puffs/L
3R4F
pMRTP
-log10(fdr p-value)
Log2 (Fold Change)

Figure 3: Volcano plots representing global differentially expressed genes (light blue dots: up-regulated; yellow dots: down-regulated; *dark blue dots: below fdr p-value of 0.05*) from NHBE cells cultures exposed for 4h to different doses of 3R4F or pMRTP smoke fractions (GVP, sbPBS, TPM) versus controls. Since the high dose of 3R4F TPM fraction induced cytotoxicity, no gene expression data is available for this condition. Abbreviations: fdr = false discovery rate.

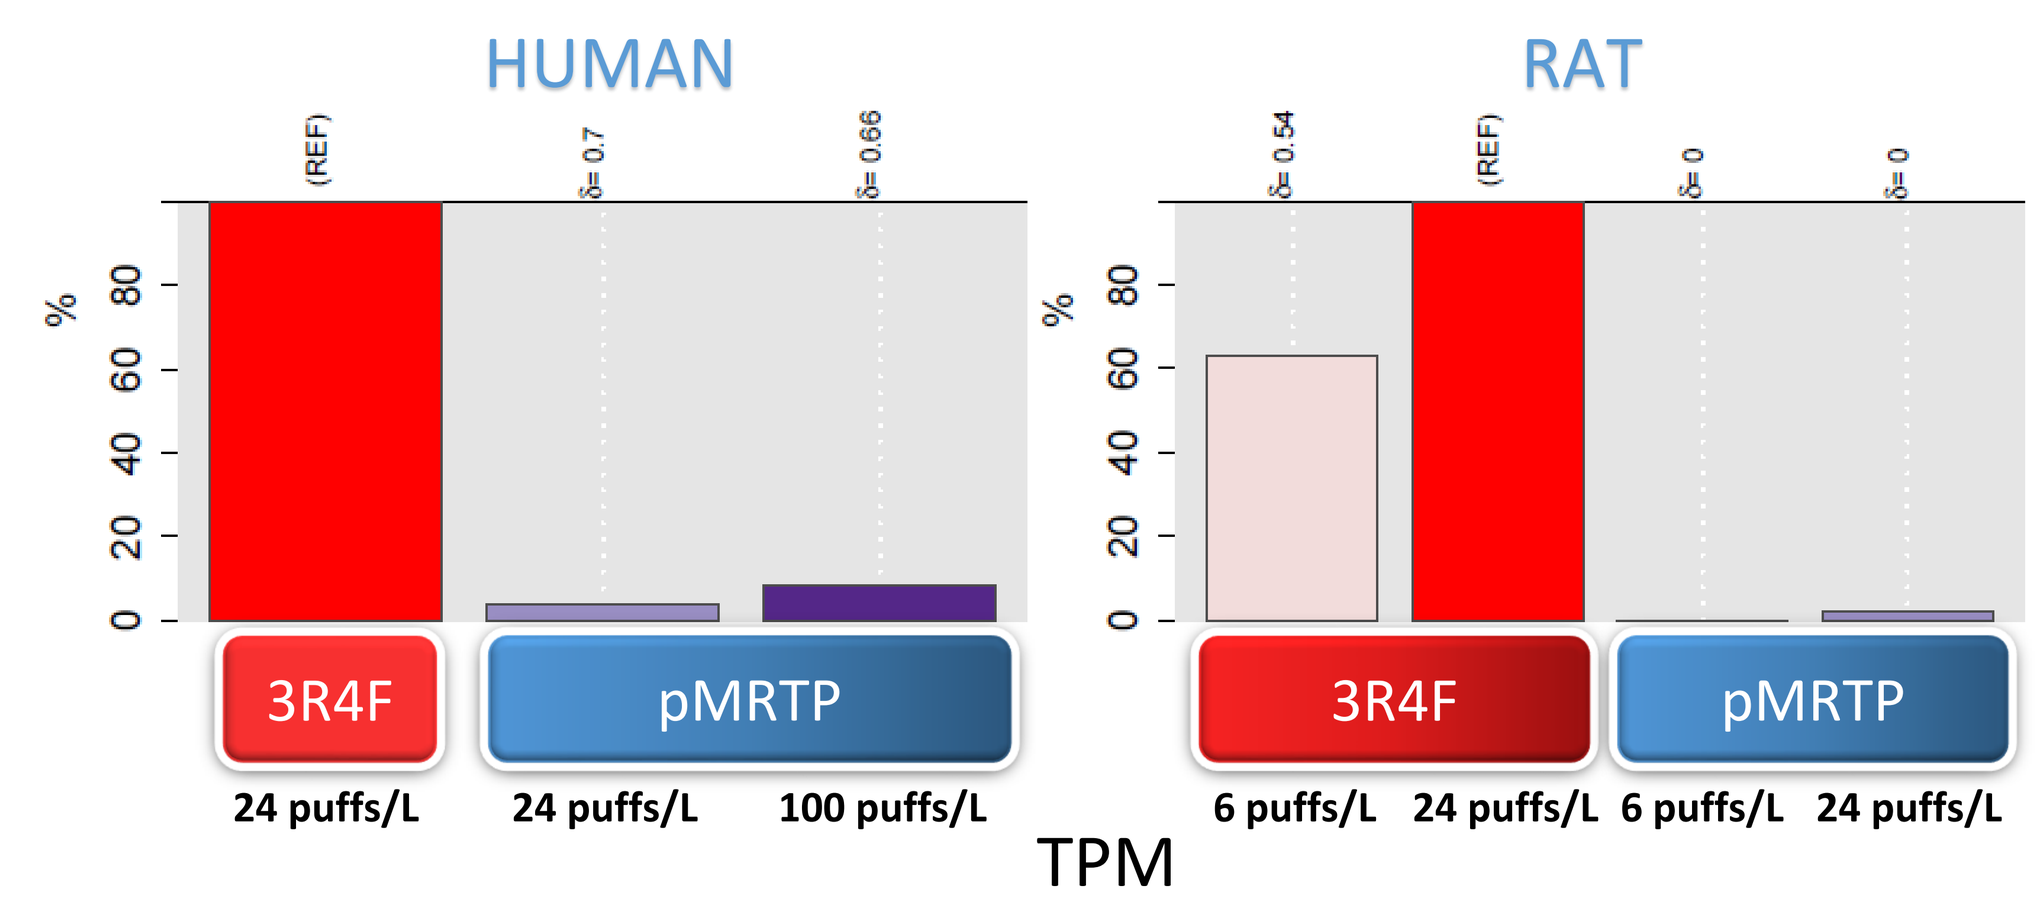
HUMAN
RAT
(REF)
δ= 0.7
δ= 0.66
δ= 0.54
δ= 0
%
3R4F
pMRTP
24 puffs/L
100 puffs/L
6 puffs/L
TPM

Figure 4: Relative BIF[7] computed for both human and rat bronchial epithelial cells exposed for 4h to different doses of TPM obtained from 3R4F smoke or pMRTP aerosol. (REF) corresponds to the system response profile for which the highest perturbation scores are observed. The delta (δ, [-1,1]) value reflects how much is the underlying perturbed biology modeled in the networks similar to the reference.

A common reduced impact of pMRTP versus 3R4F TPM but a different pattern of biological perturbations between rat and human cells

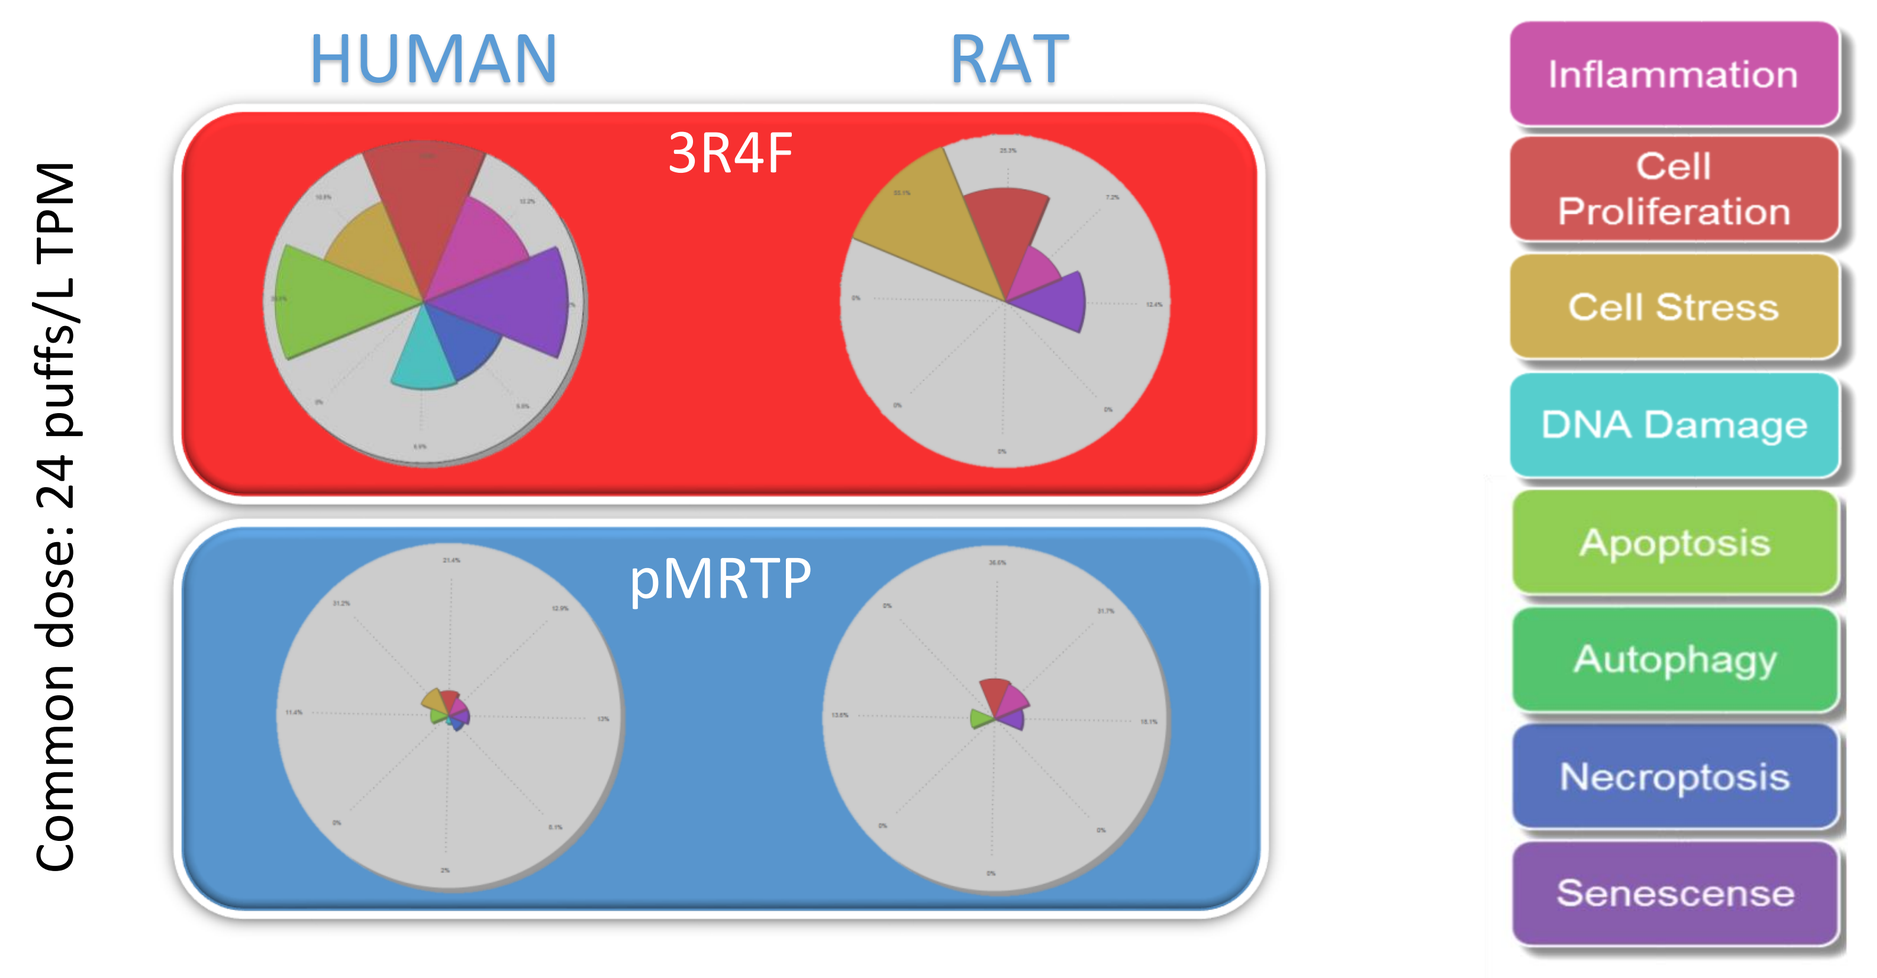
HUMAN
RAT
3R4F
pMRTP
Common dose: 24 puffs/L TPM
Inflammation
Cell Proliferation
Cell Stress
DNA Damage
Apoptosis
Autophagy
Necroptosis
Senescense

Figure 5: Under the same condition, rat and human bronchial epithelial cells were exposed for 4h to 24 puffs/L TPM from 3R4F or pMRTP smoke. The biological effect triggered in both cell types were quantified by measuring the perturbations of different biological networks (indicated in the rainbow colors boxes on the left) using gene fold-changes as an input and the NPA method[4] as a computational approach.

Correlations between the differential network backbone values in response to 3R4F TPM exposure generated from the NRBE and NHBE cells datasets in the context of causal biological network models

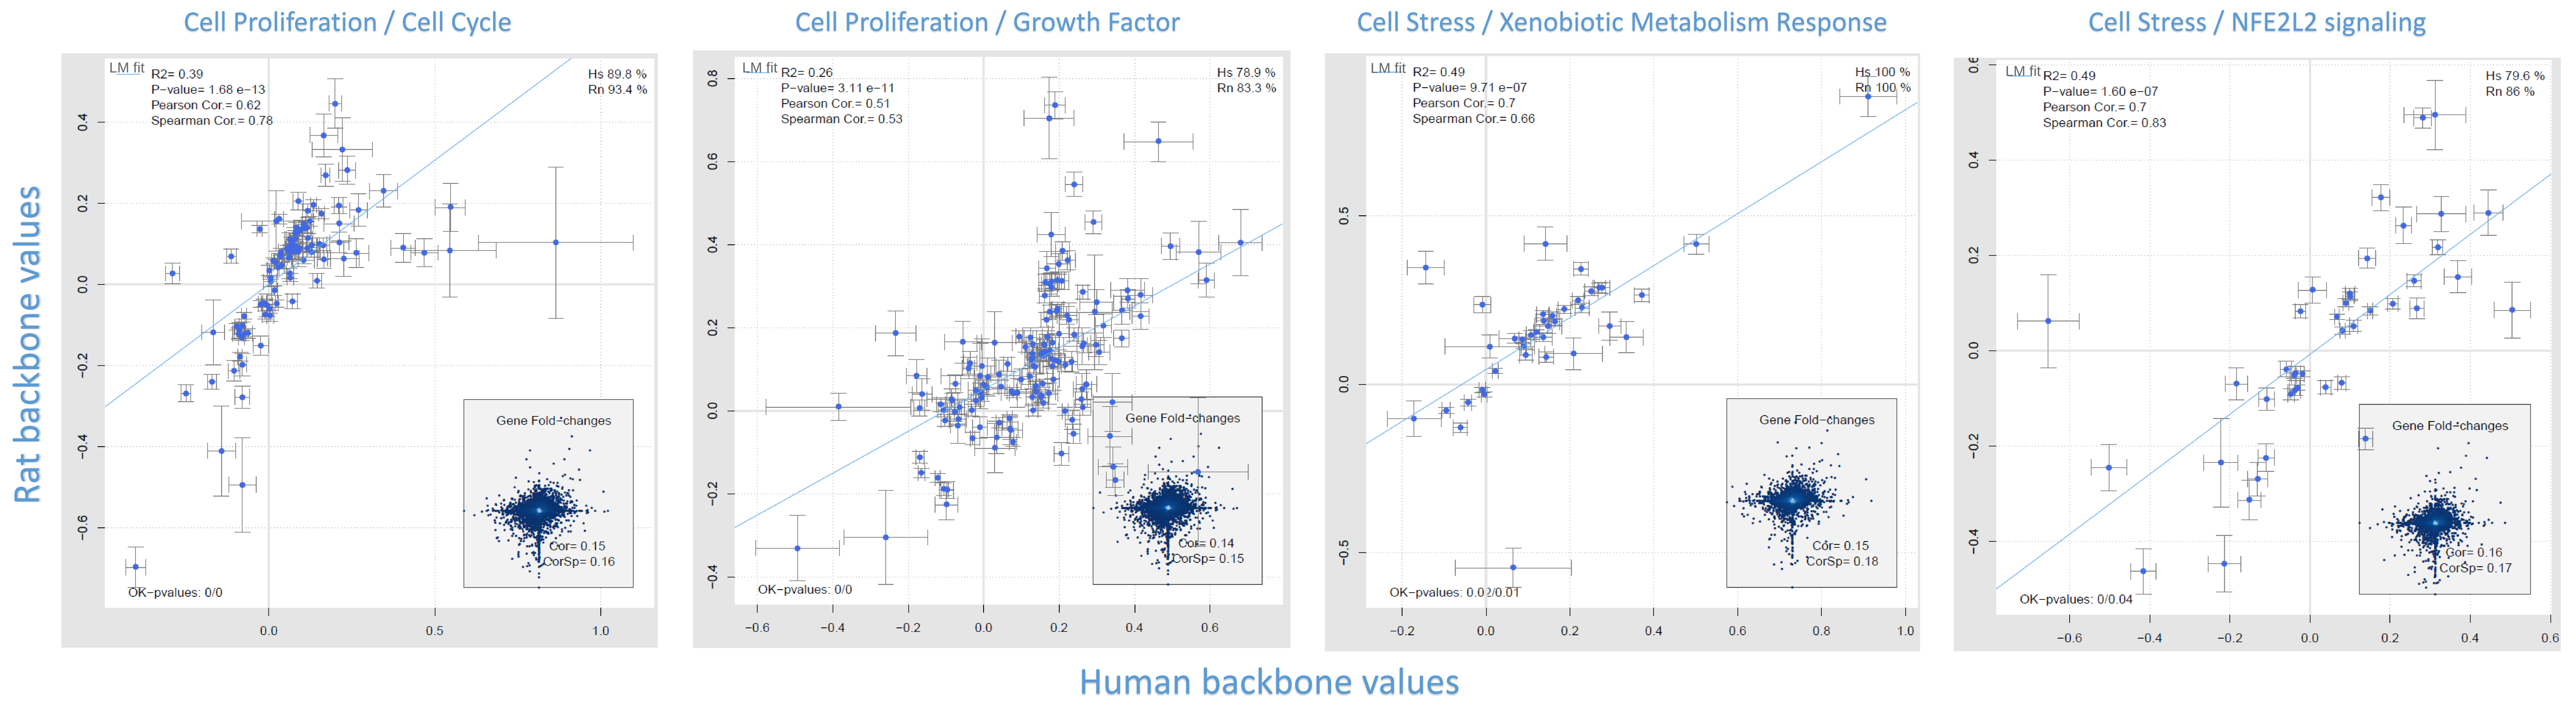
Cell Proliferation / Cell Cycle
Cell Proliferation / Growth Factor
Cell Stress / Xenobiotic Metabolism Response
Cell Stress / NFE2L2 signaling
Rat backbone values
Human backbone values
LM fit
R2= 0.39
P−value= 1.68 e−13
Pearson Cor.= 0.62
Spearman Cor.= 0.78
Hs 89.8 %
Rn 93.4 %
Gene Fold−changes
Cor= 0.15
CorSp= 0.16
OK−pvalues: 0/0
R2= 0.26
P−value= 3.11 e−11
Pearson Cor.= 0.51
Spearman Cor.= 0.53
Hs 78.9 %
Rn 83.3 %
Cor= 0.14
CorSp= 0.15
OK−pvalues: 0/0
R2= 0.49
P−value= 9.71 e−07
Pearson Cor.= 0.7
Spearman Cor.= 0.66
Hs 100 %
Rn 100 %
Cor= 0.15
CorSp= 0.18
OK−pvalues: 0.02/0.01
R2= 0.49
P−value= 1.60 e−07
Pearson Cor.= 0.7
Spearman Cor.= 0.83
Hs 79.6 %
Rn 86 %
Cor= 0.16
CorSp= 0.17
OK−pvalues: 0/0.04

Figure 6: Scatter plots of backbone values (main graph) and gene expression fold-changes (insets) on the intersection of the human and rat «Cell Cycle» (A), «Growth Factor» (B), «Xenobiotic Metabolism Response» (C) and «NFE2L2 signaling» (D) sub-networks perturbed after 3R4F TPM exposure (24 puffs/L). The upper right legend indicates the percentage of nodes in the intersection for human and rat sub-network, respectively. OK – p-values correspond to the two statistics computed to assess the significance of the network perturbation with respect to its specificity to the given two-layer network structure. R2 corresponds to the coefficient of determination in relation to the linear regression fit (LM fit). Blue lines show the linear regression lines computed by least squares fit. Cor. : Correlation. For gene expression fold changes, Cor and CorSp correspond to Pearson and Spearman correlations, respectively. Horizontal and vertical error bars correspond to the 95%-confidence interval of each human and rat backbone node (blue dot) score, respectively.

Conclusions

- NHBE cells exposed to different fractions of pMRTP showed less changes in gene expression compared to 3R4F fractions (Fig.3). A similar observation was found in rat cells (Data not shown).
- From the three smoke fractions, the most potent one from either 3R4F or pMRTP to induce gene expression changes is TPM in both rat and human cells (when comparing the same dose) (Fig. 3).
- A dose-dependent effect was observed at the gene expression level for all fractions tested for both test items (3R4F/pMRTP) in both NHBE and NRBE cells exposed (Fig. 3). For 3R4F TPM fraction, the highest dose tested (100 puffs /L) induced a strong cytotoxic response in NHBE cells, preventing gene expression analysis.
- NRBE cells were more sensitive to smoke fractions exposure compared to NHBE cells (cytotoxicity assessed by Resazurin assay, data not shown), thus only one dose in common (24 puffs/L) was investigated to compare the effect across species.
- BIF scores representing the overall quantified impact on the biological networks are presented in Figure 4 for both human and rat cells exposed to different doses of TPM. For both species, the quantified effect of pMRTP's TPM exposure is highly reduced compared to reference cigarette's TPM. The dose-dependent effect observed at the level of gene expression changes (Fig. 3) is also shown at the BIF level for both test items, in both cell types (Fig.4).
- When investigating the perturbation amplitude scores for each biological networks (Fig.5), for both species a common reduced impact of pMRTP fraction was observed when compared to 3R4F TPM.
- When comparing the effect of 3R4F TPM (24 puffs/L) exposure in both human and rat cells, differences in the cellular response clearly appear: (i) in NHBE cells, all networks were perturbed except the "Autophagy" network; (ii) in NRBE cells, only four networks were perturbed (Cell Stress, Cell Proliferation, Inflammation, Senescence); (iii) In NHBE cells, the highest NPA score was observed in the "Cell Proliferation" network while in NRBE cells, it was the "Cell Stress" network.
- In Figure 6, four significantly perturbed sub-networks ("Cell Cycle", "Growth Factor", Xenobiotic Metabolism Response" and NFE2L2 signaling") with a high percentage of nodes overlaps in both species were used to compare the species effect triggered by 3R4F TPM exposure. Comparison at the level of the backbone values showed a significant correlation between NRBE and NHBE cell responses after 3R4F TPM exposure, suggesting a similar biological effect, which was not obvious at the level of gene expression fold changes (Insets). These results suggest that certain biological processes such as stress responses or cell cycle regulation are translatable on a mechanistic level but not on a gene to gene comparison.
- In conclusion, our quantitative systems toxicology approach utilizing causal network models representing key biological mechanisms is a powerful tool to analyze and compare the effect of different fractions from conventional cigarette or from Modified Risk Tobacco Products. A better understanding of the range of applicability of the translation concept will impact the predictability of signaling responses, mode of action and efficacy of drugs in the field of systems pharmacology as well as increase the confidence in the estimation of human risk from rodent data in the context of toxicological risk assessment.

References

1. **A network-based approach to quantifying the impact of biologically active substances.** *J. Hoeng, et al.* Drug Discovery Today, 2012 May 1; 28(9):1193-201.
2. **Construction of a computable cell proliferation network for non-diseased lung tissue.** *J. Westra, et al.* BMC Systems Biology, 2011 Jul 5:105.
3. **Construction of a computable cellular stress network for non-diseased lung and cardiovascular tissue.** *W.K. Schlage, et al.* BMC Systems Biology, 2011 Oct 5:168.
4. **Assessment of network perturbation amplitude by applying high-throughput data to causal biological networks.** *F. Martin, et al.* BMC Systems Biology 2012, 6:54.
5. **Construction of a Computable Network Model for DNA Damage, Cell Death, Autophagy, and Senescence.** *S. Gebel, et al.* Bioinformatics and Biology Insights 2013 7:97-117.
6. **A modular cell-type focused inflammatory process network model for non-diseased pulmonary tissue.** *J. Westra, et al.* Bioinformatics and Biology Insights 2013 Jun 20; 7:167-92.
7. **Quantification of biological network perturbations for mechanistic insight and diagnostics using two-layer causal models assessment of biological impact using transcriptomic data and mechanistic network models.** *F. Martin, et al.* BMC Bioinformatics. 2014 Jul 11;15(1):238.

The research described here was supported by Philip Morris International.

PMI RESEARCH & DEVELOPMENT

Eurotox EDINBURGH 2014 — 50th Congress of the European Societies of Toxicology, 7th-10th September 2014 • EICC Edinburgh

Philip Morris International Research & Development, Quai Jeanrenaud 5, 2000 Neuchâtel, Switzerland
T: +41 58 242 21 11, **F**: +41 58 242 28 11, **W**: www.pmi.com